Real-World Treatment Preferences Among People Living with ALS: A Discrete Choice Experiment

PCR162

Katie Stenson[1], Lara Taylor[1], Nicholas Belviso[1], Teresa E. Fecteau[1], Paula Alvarez[1], Nandini Hadker[2], Matthew O'Hara[2], Amod Athavale[2], Olivia Green[2], Vijay Abilash[2], Lucas Monserrat[2], Cali Orsulak,[3] Raquel Norel,[4,5] Meera Gandhi [5]

[1]Biogen, 225 Binney Street, Cambridge, MA, USA, [2]Trinity Life Sciences, Waltham, MA, USA, [3]Northeast Amyotrophic Lateral Sclerosis Consortium, MA, USA, [4]IBM Research, Yorktown Heights, NY, USA, [5]EverythingALS, Seattle, WA, USA

Objective

Quantitatively assess which treatment attributes are most important to people living with amyotrophic lateral sclerosis (ALS; pALS) in the United States (US) when making treatment decisions

Conclusions

- Through direct and indirect assessment of preference, pALS indicated a desire for efficacious treatment options that improve physical functioning and survival. pALS showed a willingness to trade off attributes like ROA, and flexibility on monitoring requirements if it resulted in greater safety
- Disconnect seen in the DCE results as pALS highly ranked the importance of non-targeted therapy, but the efficacy and safety attribute levels preferred are currently only achievable through targeted therapies. This disparity is thought to be generated by a desire for the broad ALS community to have access to efficacious therapies, and not only a subset of the population. High importance may also be impacted by high proportion of the sample likely being genotype-negative and not currently being eligible for targeted therapies
- Results highlight the continued unmet need for more efficacious therapies in ALS, with 69% of pALS reporting a need to leave their current job/school due to their ALS and a high impact of ALS on quality of life, especially due to difficulty walking, speaking and holding objects
- These results will allow for the patient's treatment attribute preferences to be better incorporated into physician/patient treatment decision-making and into further ALS drug development

Limitations

- Multi-model recruitment techniques (patient panels, via treating physician, patient advocacy groups, and social media outreach) were used to drive recruitment, which might have introduced a sampling bias (56% responses generated from PAGs)
- DCE study design and analysis is complex – the number of included treatment attributes was limited due to sample size and to reduce patient burden
- Questions about events in the past may have been subject to recall bias
- Data provided by participants were self-reported and not verified by a physician

Introduction

- ALS is a rare, progressive neurodegenerative disease characterized by damage and loss of both upper and lower motor neurons, which commonly presents as progressive muscle weakness. The majority of pALS experience death from respiratory failure within the first 3-5 years[1-4]
- There are three treatments currently approved for use in ALS (riluzole, edaravone, and sodium phenylbutyrate & taurursodiol) for pALS regardless of genetic subtype[5]
- New agents that target specific genetic mutations associated with ALS are in development. Tofersen is a genetically targeted therapy for *SOD1*-ALS which is undergoing FDA review, with a PDUFA date of April 25, 2023[6-7]

Methods

- A 30-minute, Institutional Review Board (IRB)-approved, web-enabled questionnaire, including a discrete choice experiment (DCE), was fielded between October 2022 and January 2023 among pALS and caregivers of pALS responding as proxies for patients residing in the US
- Pretest interviews (n=4) were conducted to test questionnaire understanding by participants and obtain qualitative feedback
- In the DCE, participants were presented with various product configurations representing attributes from ALS treatment options that are currently available or expected to enter the market by 2026. Some attributes of the treatment profiles were not directly related to trial data and were aspirational
- A 15-choice-set experimental design with 3 hypothetical treatment profiles in each choice set was created and the data were analyzed using a hierarchical Bayesian model
- Participants were asked to select their most preferred product from each choice set and could select a 'none' option if they preferred their current ALS treatment regimen over the product configurations presented in the choice set
- ALS Milano-Torino staging system (MiToS) was used to ascertain the ALS disease severity of patients based on loss of function in four key domains (walking or self-care, swallowing, communicating, and breathing) in the Revised ALS Functional Rating (ALSFRS-R scale), a higher MiToS score corresponds to greater disease severity[8-9]

TABLE 1. Inclusion and Exclusion Criteria of the Study Participants

Inclusion Criteria	Exclusion Criteria
≥21 years of age	Current employees of or affiliation with the following:
Individuals who have been told by a physician that they have ALS	• Pharmaceutical manufacturers, biotech, or medical equipment manufacturer
Open to trying a new drug for ALS if a physician recommended it (rated on 1-9 scale with 1 = "not open at all" and 9 = "extremely open"; only included if response was ≥3)*	• Contract research organization • Market research or advertising firm • FDA or government agency

Note: *A total of n=2 individuals provided a score < 3, and those individuals were excluded from the study

TABLE 2. ALS Discrete Choice Experiment Design Grid

Attribute	Level 1	Level 2	Level 3
Type of Therapy	Product that works for all pALS; not for specific genetic types of ALS (non-targeted)	Product that works for a specific genetic type of ALS (targeted)	--
Route of Administration (ROA)	Intrathecal given once monthly	Oral formulation twice daily	Oral formulation once daily
Effect of Product on Neurofilament Levels in Clinical Study	No impact on neurofilament	Early and lasting neurofilament reduction	--
Effect of Product on the ALSFRS-R Measuring Disease Progression / Physical Function in Clinical Study	2.5-point difference in disease progression / physical function observed after 6 months compared to study participants not on Product	3.5-point difference in disease progression / physical function observed after 1 year compared to study participants who had a 6-month delay on Product	6.0-point difference in disease progression / physical function observed after 1 year compared to study participants not on Product
Effect of Product on Survival in Clinical Study	No impact on survival was shown for the study participants on this Product	Estimated decreased risk of death / permanent ventilation by 40% after 6 months compared to study participants not on Product	Estimated decreased risk of death or permanent ventilation by 65% after 1.5 years compared to study participants who had a 6-month delay on Product
Efficacy of Product on Lung Function in Clinical Study	Slower worsening of lung function with an average difference of 4 points in percent predicted vital capacity at 9 months compared to participants not on Product	Slower worsening of lung function with an average difference of 5 points in percent predicted vital capacity at 6 months compared to participants not on Product	Slower worsening of lung function with an average difference of 9 points in percent predicted vital capacity at 1 year compared to study participants who had a 6-month delay on Product
Safety	Cardiac events / ECG abnormalities (15%)	Serious neurological events (7%)	No serious adverse events were observed
Monitoring Required for Product Use	Blood and urine monitoring prior to each monthly dose	After the first dose, monitoring of signs and symptoms of product-related side effects during visits	No side effect-related monitoring required
Genetic Testing Required for Product Use*	Genetic testing required	No genetic testing is required	--

Note: *Genetic testing attribute was restricted with type of therapy attribute – targeted therapy was only shown with 'genetic testing required' to receive product, Non-targeted was only shown with 'no genetic testing required'

Results

TABLE 3. Respondent Demographics

pALS Characteristics	N=88
Participant Type [n (%)]	
• pALS	80 (90.9%)
• Caregiver proxies of pALS	8 (9.1%)
Age [mean (SD)]	51.1 (13.7) years
Time Since ALS Symptom Onset [mean (SD)]	5.1 (4.4) years
Time Since Told They have ALS (by physician) [mean (SD)]	4.1 (4.9) years
Number of Physicians Seen Prior to Being Told They Have ALS [mean (SD)]	3.5 (1.7)
Type of Physician pALS are Most Often Treated By Currently [mean (SD)]	
• ALS specialist	52.3%
• Neurologist	33.0%
• General Practitioner / primary care physician	13.6%
• Other	1.1%
Undergone Genetic Testing (%)	75.0%
Race/Ethnicity Distribution (%)	
• Non-Hispanic White	86.4%
• Non-Hispanic Black or African American	8.0%
• Hispanic or Latino	3.4%
• Native American or American Indian	1.1%
• Other	1.1%
Health Insurance Coverage Distribution (%)	
• Medicare	70.5%
• Employer provided private/ commercial insurance	31.8%
• Medicaid	19.3%
• Self-purchased private/ commercial insurance	12.5%
• Other government insurance (i.e., VA, DOD)	2.3%
• Patient assistance	2.3%
Dual coverage possible so percentages exceed 100%	
pALS Disease Severity Segmentation Based on MiToS Stage [n(%)]	
• Stage 0	41 (46.6%)
• Stage 1	29 (33.0%)
• Stage 2	12 (13.6%)
• Stage 3	2 (2.3%)
• Stage 4	4 (4.5%)
Impact ALS Has Had on Lives of pALS	
• Negative impact on familial and social wellbeing	73 (83%)
• Need to leave current job / school	61 (69%)
• Need to reduce responsibilities at work / go to school part time	52 (59%)
• Turned down an opportunity at job / school	48 (55%)

FIGURE 1. Direct Assessment: ALS Treatment Utilization – Overall Sample (N=88)

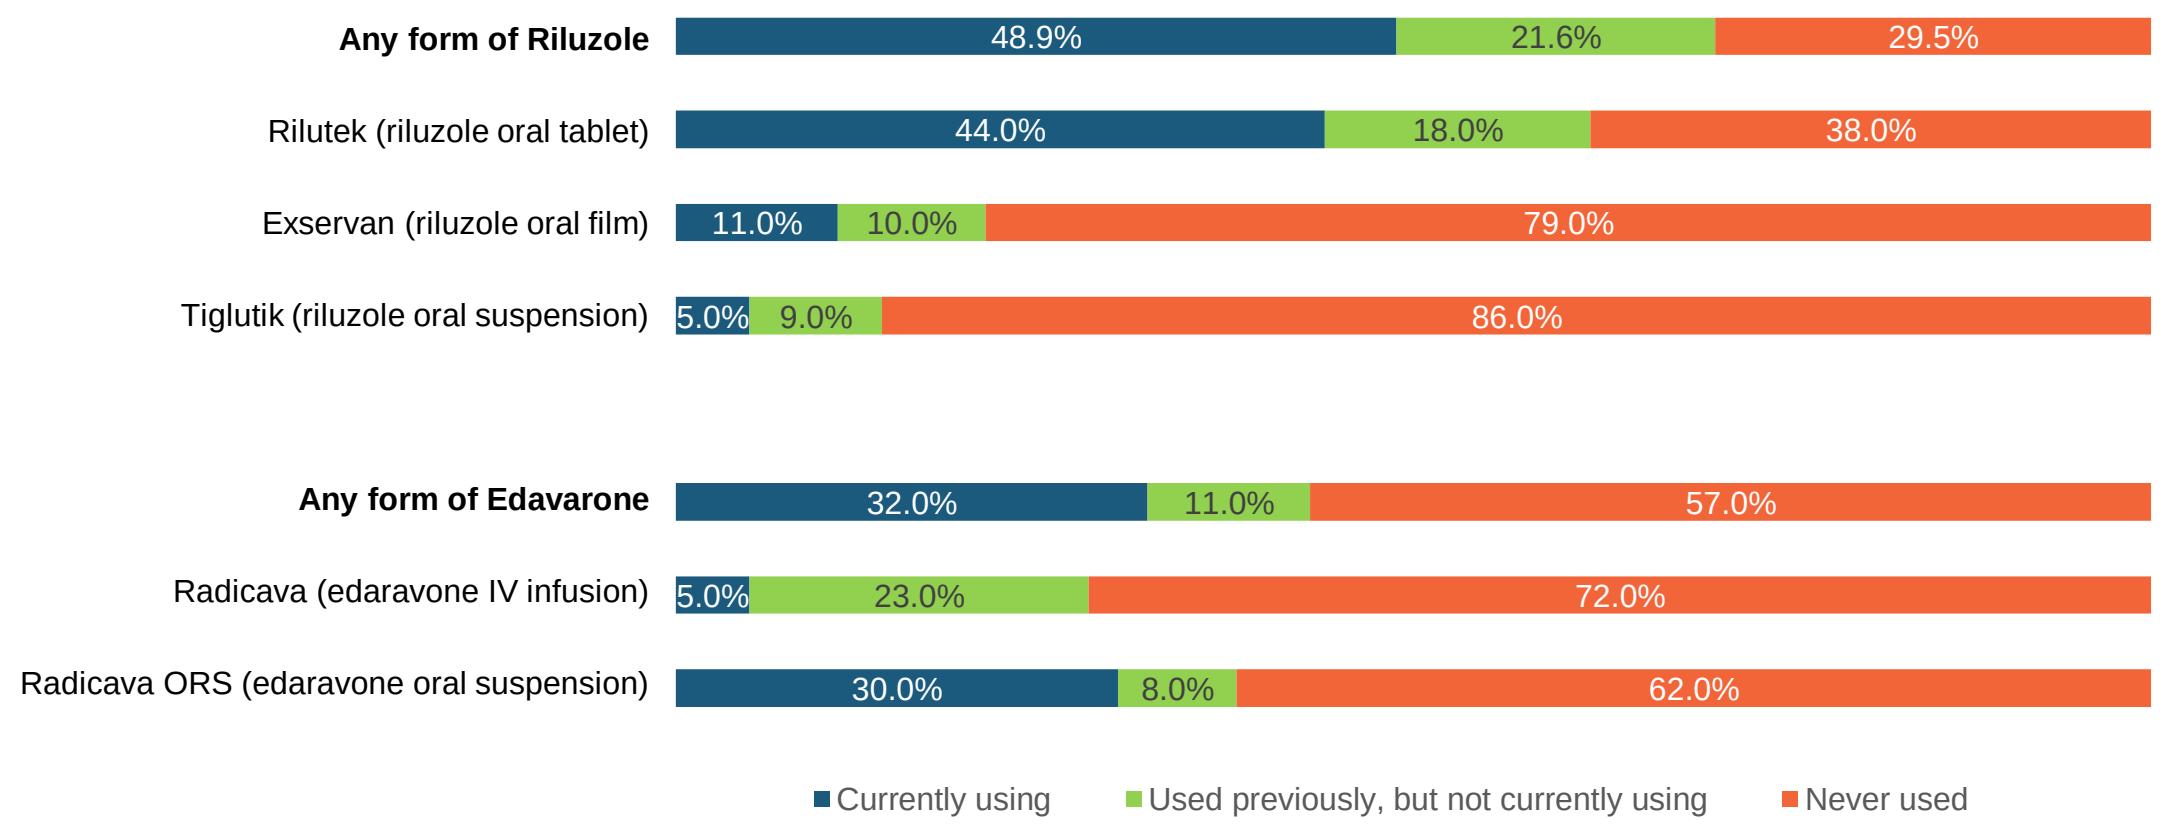
Any form of Riluzole
48.9%
21.6%
29.5%
Rilutek (riluzole oral tablet)
44.0%
18.0%
38.0%
Exservan (riluzole oral film)
11.0%
10.0%
79.0%
Tiglutik (riluzole oral suspension)
5.0%
9.0%
86.0%
Any form of Edavarone
32.0%
11.0%
57.0%
Radicava (edaravone IV infusion)
5.0%
23.0%
72.0%
Radicava ORS (edaravone oral suspension)
30.0%
8.0%
62.0%
Currently using
Used previously, but not currently using
Never used

Note: ~65% of pALS who have previously taken and discontinued Radicava IV infusion have since switched to Radicava ORS. Sodium phenylbutyrate & taurursodiol is not included here as regulatory approval was received after study initiation

FIGURE 2. DCE Output: Mean Relative Attribute Importance – Overall Sample (N=88)

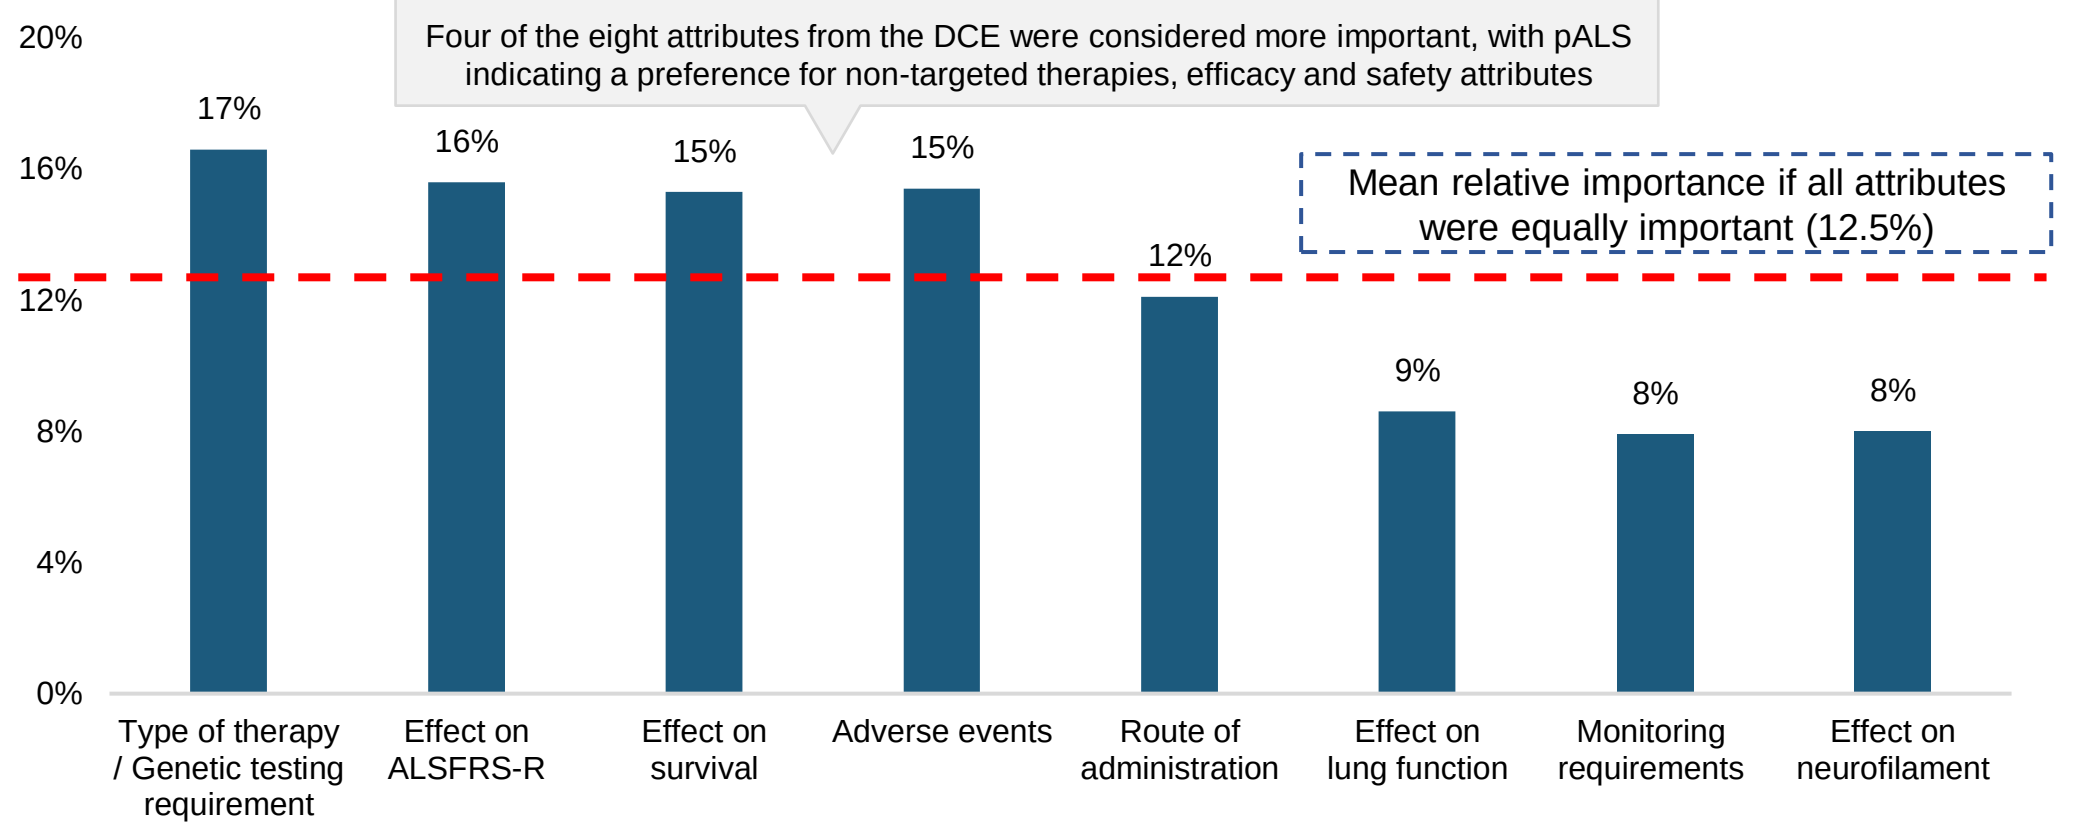
Four of the eight attributes from the DCE were considered more important, with pALS indicating a preference for non-targeted therapies, efficacy and safety attributes
Mean relative importance if all attributes were equally important (12.5%)
20%
16%
12%
8%
4%
0%
17%
16%
15%
15%
12%
9%
8%
8%
Type of therapy / Genetic testing requirement
Effect on ALSFRS-R
Effect on survival
Adverse events
Route of administration
Effect on lung function
Monitoring requirements
Effect on neurofilament

Note: Genetic testing requirement was restricted to the type of therapy as the two attributes are perfectly correlated and testing separately would have resulted in a flawed DCE design

FIGURE 3. Direct Assessment: Mean Product Attribute Importance when pALS Consider an ALS Treatment – Overall Sample (N=88)

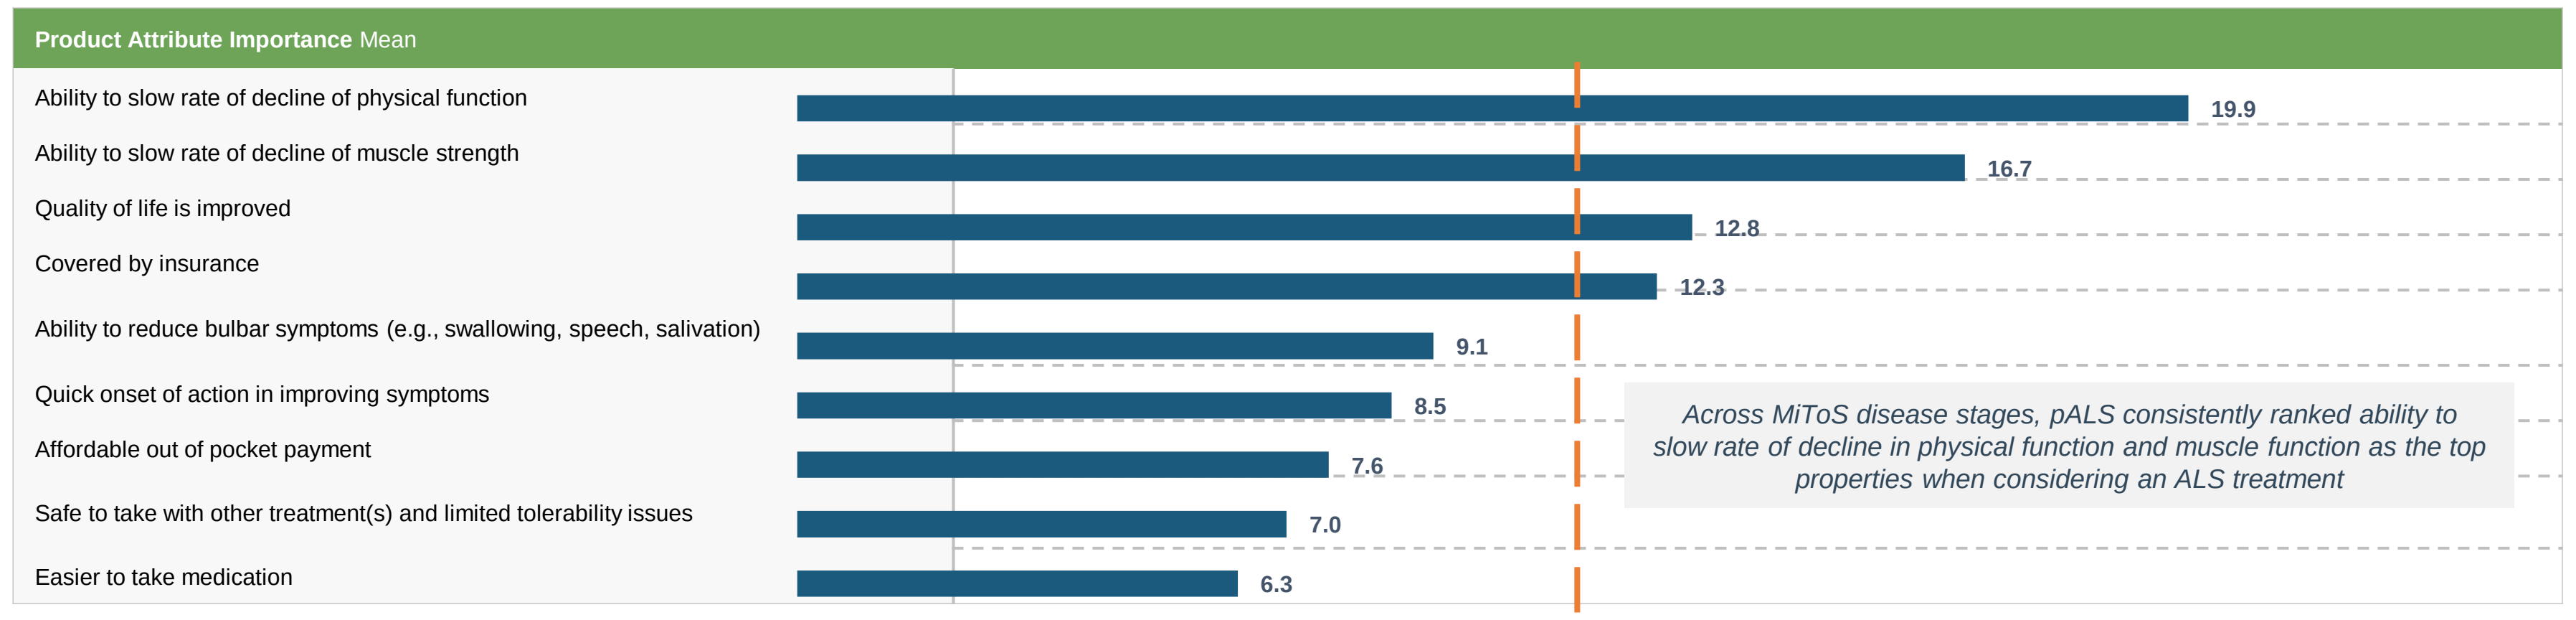
Product Attribute Importance Mean
Ability to slow rate of decline of physical function
19.9
Ability to slow rate of decline of muscle strength
16.7
Quality of life is improved
12.8
Covered by insurance
12.3
Ability to reduce bulbar symptoms (e.g., swallowing, speech, salivation)
9.1
Quick onset of action in improving symptoms
8.5
Affordable out of pocket payment
7.6
Safe to take with other treatment(s) and limited tolerability issues
7.0
Easier to take medication
6.3
Across MiToS disease stages, pALS consistently ranked ability to slow rate of decline in physical function and muscle function as the top properties when considering an ALS treatment

Note: pALS asked to allocate 100 points across each attribute with higher point allocations corresponding to greater importance relative to other attributes; N=88). Attributes with ratings higher than 11 (100 possible points to allocate divided by 9 attributes tested) were considered more important compared to other attributes.

*pALS indicated that quality of life would be most improved by addressing difficulty walking, speaking and holding objects

FIGURE 4. Direct Assessment: Attitudinal Responses – Overall Sample (N=88)

Completely Disagree 1 2 3 4 5 6 7 8 9 *Completely Agree*

In order of increasing average agreement

Attitudinal	Mean Agreement Rating	% Reporting a Rating of ≥7
Valuable if a product can stabilize my ALS symptoms	7.8	77%
Will take an ALS treatment with greater side effect risk if it is targeted for my specific ALS genetic mutation	7.3	72%
Will take an ALS treatment with an intrathecal ROA if a product stabilizes my ALS symptoms	6.9	65%
Open to genetic testing to take an ALS treatment that is targeted for my specific ALS genetic mutation	6.7	60%

Note: Among pALS who had undergone genetic testing (GT), 65% selected 7 or more (on a 1-9 scale, where 9 is 'completely agree') on the statement 'Open to undergoing GT to take an ALS treatment option that is targeted for specific ALS genetic mutation'. Even among the pALS who haven't undergone GT, 45% of them have selected 7 or more on the 'open to GT' statement.

For pALS who had not undergone GT, 68% indicated that physician didn't bring up the need for testing, followed by 14% indicating concerns around misuse of results, and 14% indicating insurance coverage issues

Acknowledgments and Disclosure

Medical writing support was provided by Bhagyashree Oak, PhD and Latashree Goswami, MPH of Trinity Life Sciences. Funding for this study was provided by Biogen

References

1. Brown RH, Al-Chalabi A. Amyotrophic Lateral Sclerosis. *N Engl J Med*. 2017;377(2):162-172.
2. Al-Chalabi A, Hardiman O. The epidemiology of ALS: a conspiracy of genes, environment and time. *Nat Rev Neurol.* 2013;9(11):617-628. doi:10.1038/nrneurol.2013.203
3. Brown RH, Al-Chalabi A. Amyotrophic Lateral Sclerosis. *N Engl J Med*. 2017;377(2):162-172. doi:10.1056/NEJMra1603471
4. Bunton-Stasyshyn RK, Saccon RA, Fratta P, Fisher EM. SOD1 Function and Its Implications for Amyotrophic Lateral Sclerosis Pathology: New and Renascent Themes. *Neuroscientist*. 2015;21(5):519-529. doi:10.1177/1073858414561795
5. Goyal NA, Galvez-Jimenez N, Cudkowicz ME. Disease-modifying treatment of amyotrophic lateral sclerosis. *UpToDate.* https://www.medilib.ir/uptodate/show/5128. Accessed March 15, 20123.
6. Miller TM, Cudkowicz ME, Genge A, et al. Trial of Antisense Oligonucleotide Tofersen for SOD1 ALS. *N Engl J Med.* 2022;387(12):1099-1110. doi:10.1056/NEJMoa2204705
7. FDA Accepts Biogen's New Drug Application and Grants Priority Review of Tofersen for a Rare, Genetic Form of ALS. News release. JULY 26, 2022. https://investors.biogen.com/news-releases/news-release-details/fda-accepts-biogens-new-drug-application-and-grants-priority. Accessed April 4, 2023
8. Chiò A, Hammond ER, Mora G, Bonito V, Filippini G. Development and evaluation of a clinical staging system for amyotrophic lateral sclerosis. *J Neurol Neurosurg Psychiatry.* 2015;86(1):38-44. doi:10.1136/jnnp-2013-306589
9. He, Ruojie & Zheng, Minying & Lian, Ling & Yao, Xiaoli. (2021). Milano–Torino Staging and Long-Term Survival in Chinese Patients with Amyotrophic Lateral Sclerosis. *Cells.* 10. 1220. 10.3390/cells10051220.

ISPOR 2023 | May 7–10, 2023 | Boston, MA

Reach out to katie.stenson@biogen.com with any questions