EE258

National Trends in Costs and Site of Service of Infused Disease-Modifying Therapies for Multiple Sclerosis

Elmor D Pineda, Achal Patel, Carmen Ng

Genentech, Inc., South San Francisco, CA, USA

BACKGROUND

- Intravenous (IV) disease-modifying therapies (DMT) for multiple sclerosis (MS) are subject to site-of-service markups by providers, specifically occurring in the hospital outpatient department (HOPD) setting,[1] which leads to variability in the true cost to payers
- Ocrelizumab (OCR) is the first-approved, high-efficacy DMT for both relapsing and primary progressive forms of MS and is administered by IV infusion twice yearly[2]
- Prior studies estimating the real-world cost of OCR limited their analyses to patients treated near the time of launch, thus reflecting inflated estimates of OCR costs without respect to changes over time[3]
- Natalizumab (NAT), approved in 2004, is another high-efficacy DMT for relapsing MS and is administered by IV infusion once monthly[4] or at extended intervals (every ≥6 weeks) to mitigate NAT-related risk of progressive multifocal leukoencephalopathy[5]
- Limited data exist to understand the evolution of real-world utilization and cost of OCR by site of service over time and to understand these costs compared with NAT

OBJECTIVE

- To identify and compare national trends in OCR and NAT costs and site-of-service distribution over time

METHODS

Study Design and Data Source

- This retrospective, repeated cross-sectional study of commercial claims in the US included fully adjudicated claims data from PharMetrics Plus from January 1, 2018–June 30, 2022 and assessed national quarterly trends in the real-world cost and site-of-service distribution of OCR and NAT utilization

Inclusion/Exclusion Criteria

- Patient claims with any code for OCR [Healthcare Common Procedure Coding System (HCPCS) J code (J2350, C9494) or NAT J code (J2323) or National Drug Code (NDC) for OCR (50242015001)] or NAT (6440400801) during the study period were included
- Patients who had any OCR or NAT claims (medical or pharmacy) where the allowed cost was ≤$0 at any time during the entire study period were excluded

Measures of Interest

- Number of OCR and NAT doses:
 - Presented by quarter starting from January 2018 through June 2022
 - Distribution of OCR and NAT administrations by site of service, by quarter and by year
 - Site of service: HOPD, office and other
- Cost of OCR and NAT doses:
 - Mean and median quarterly and annual OCR and NAT allowed costs, overall and by site of service
 - Mean and median annual OCR and NAT out-of-pocket (OOP) costs, overall and by site of service
 - Drug costs are measured as total allowed costs and OOP costs; costs adjusted to 2021 USD
 - Total allowed costs are the maximum amount a plan will pay for a covered healthcare service, billed under OCR HCPCS/NDC code only (no administration costs)
 - OOP costs are healthcare service expenses that are not reimbursed by a plan

OCR Loading and Maintenance and NAT Regular Doses

- For OCR, claims for loading doses were separated from maintenance doses since each loading dose is half the cost of a maintenance dose. Combining loading and maintenance dose claims would underestimate overall mean costs (**Figure 1**)
 - OCR loading dose: two claims associated with 300-mg OCR doses separated 13 to 21 days apart
 - OCR maintenance dose: claims associated with 600-mg OCR dose
 - NAT regular dose: claims associated with 300-mg NAT dose

Figure 1. Schedule of Doses for OCR and NAT

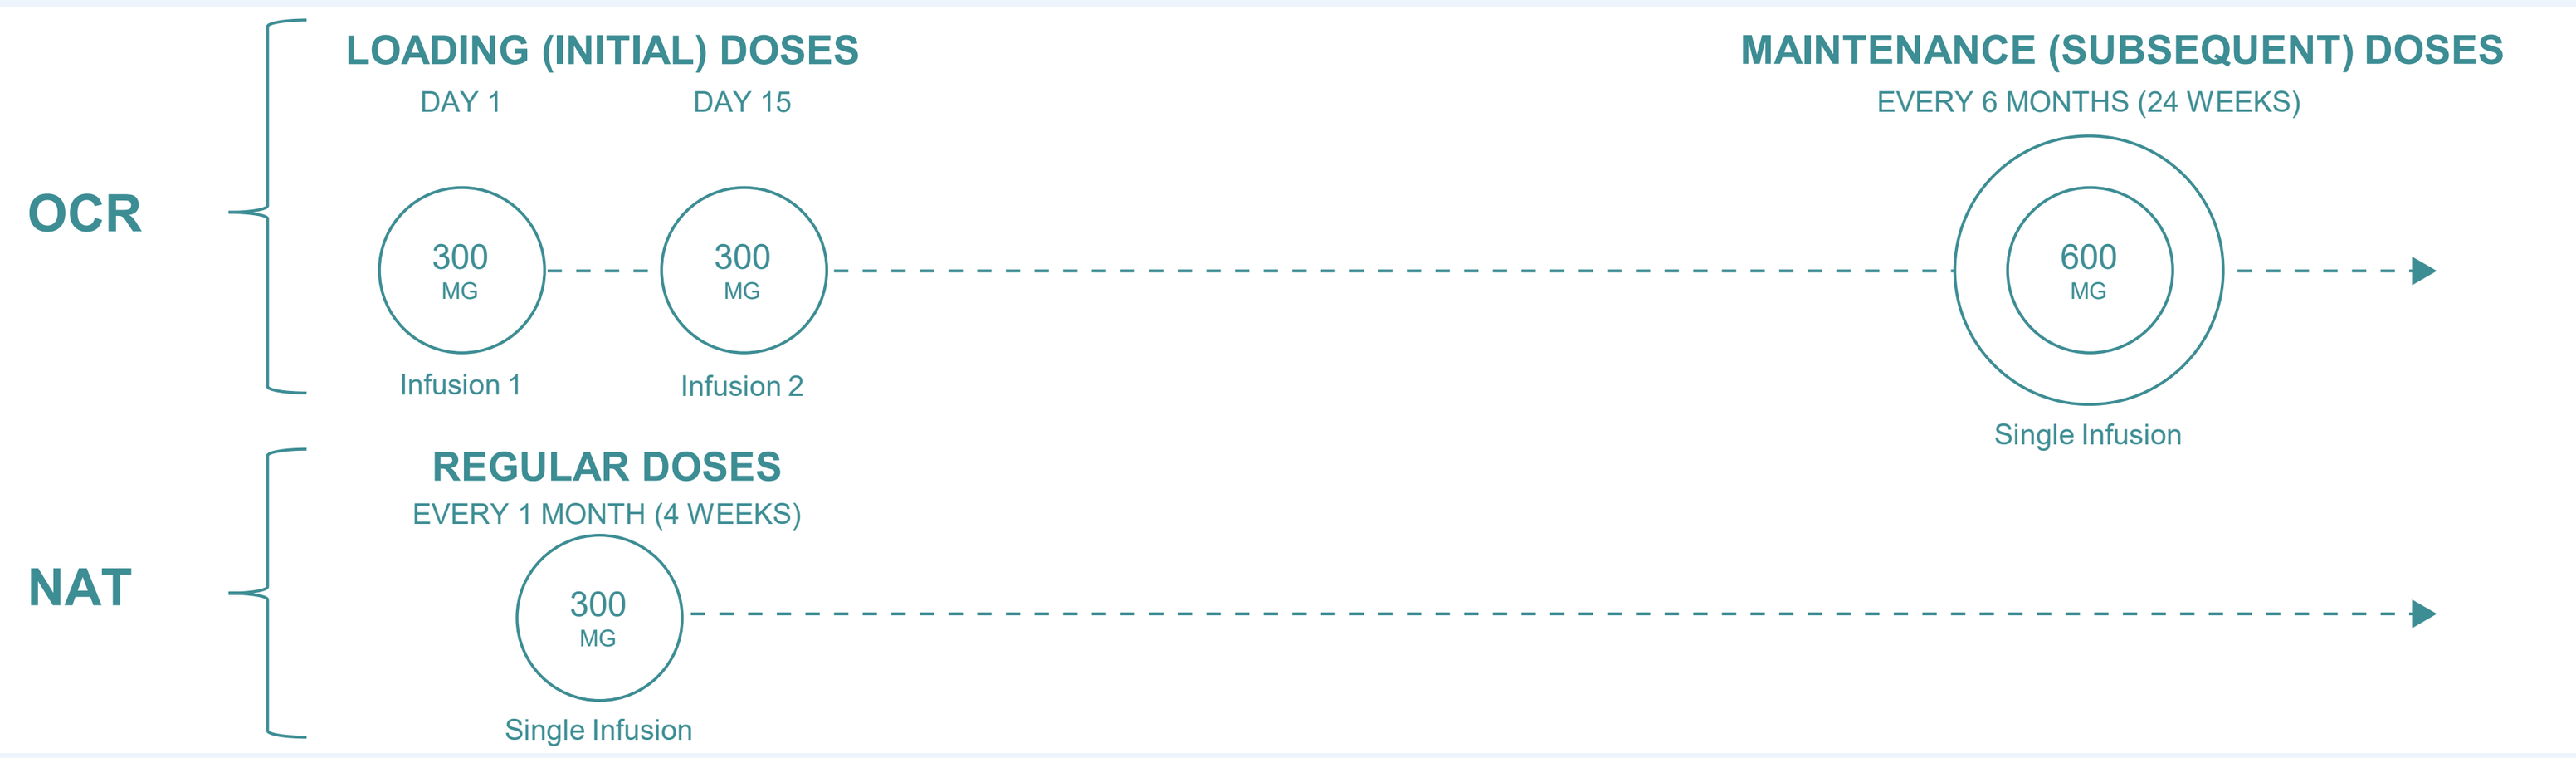
OCR
LOADING (INITIAL) DOSES
DAY 1
DAY 15
300 MG
Infusion 1
300 MG
Infusion 2
MAINTENANCE (SUBSEQUENT) DOSES
EVERY 6 MONTHS (24 WEEKS)
600 MG
Single Infusion
NAT
REGULAR DOSES
EVERY 1 MONTH (4 WEEKS)
300 MG
Single Infusion

NAT, natalizumab; OCR, ocrelizumab.

RESULTS

Figure 2. Sample Selection

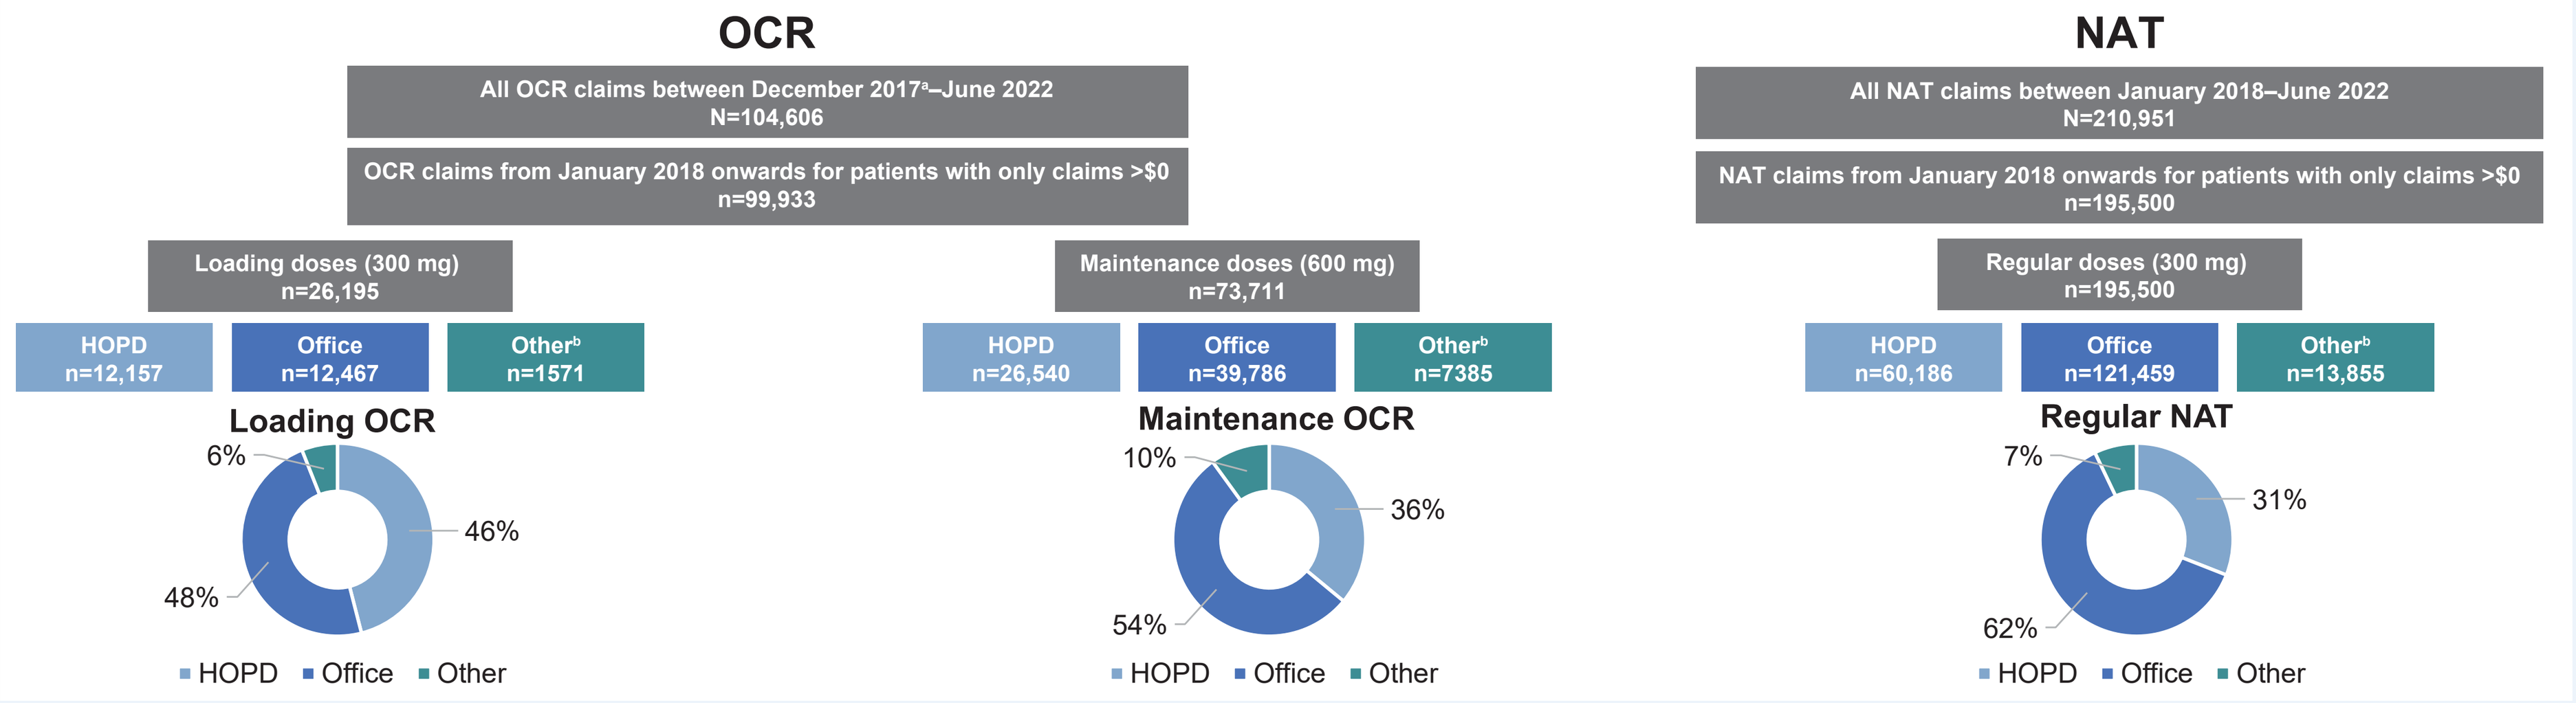
OCR
All OCR claims between December 2017ᵃ–June 2022 N=104,606
OCR claims from January 2018 onwards for patients with only claims >$0 n=99,933
Loading doses (300 mg) n=26,195
HOPD n=12,157
Office n=12,467
Otherᵇ n=1571
Loading OCR
6%
46%
48%
Maintenance doses (600 mg) n=73,711
HOPD n=26,540
Office n=39,786
Otherᵇ n=7385
Maintenance OCR
10%
36%
54%
NAT
All NAT claims between January 2018–June 2022 N=210,951
NAT claims from January 2018 onwards for patients with only claims >$0 n=195,500
Regular doses (300 mg) n=195,500
HOPD n=60,186
Office n=121,459
Otherᵇ n=13,855
Regular NAT
7%
31%
62%
HOPD
Office
Other

HOPD, hospital outpatient department; NAT, natalizumab; OCR, ocrelizumab.
[a]Data from December 2017 were used to distinguish loading vs maintenance for doses identified in January 2017.
[b]Other includes all administration sites outside of HOPD and office (pharmacy, inpatient hospital, emergency department, state/local/rural health clinics, etc).

Figure 4. Mean and Median Cost Per OCR Maintenance and NAT Regular Dose Over Time (2018 Q1–2022 Q2)[a]

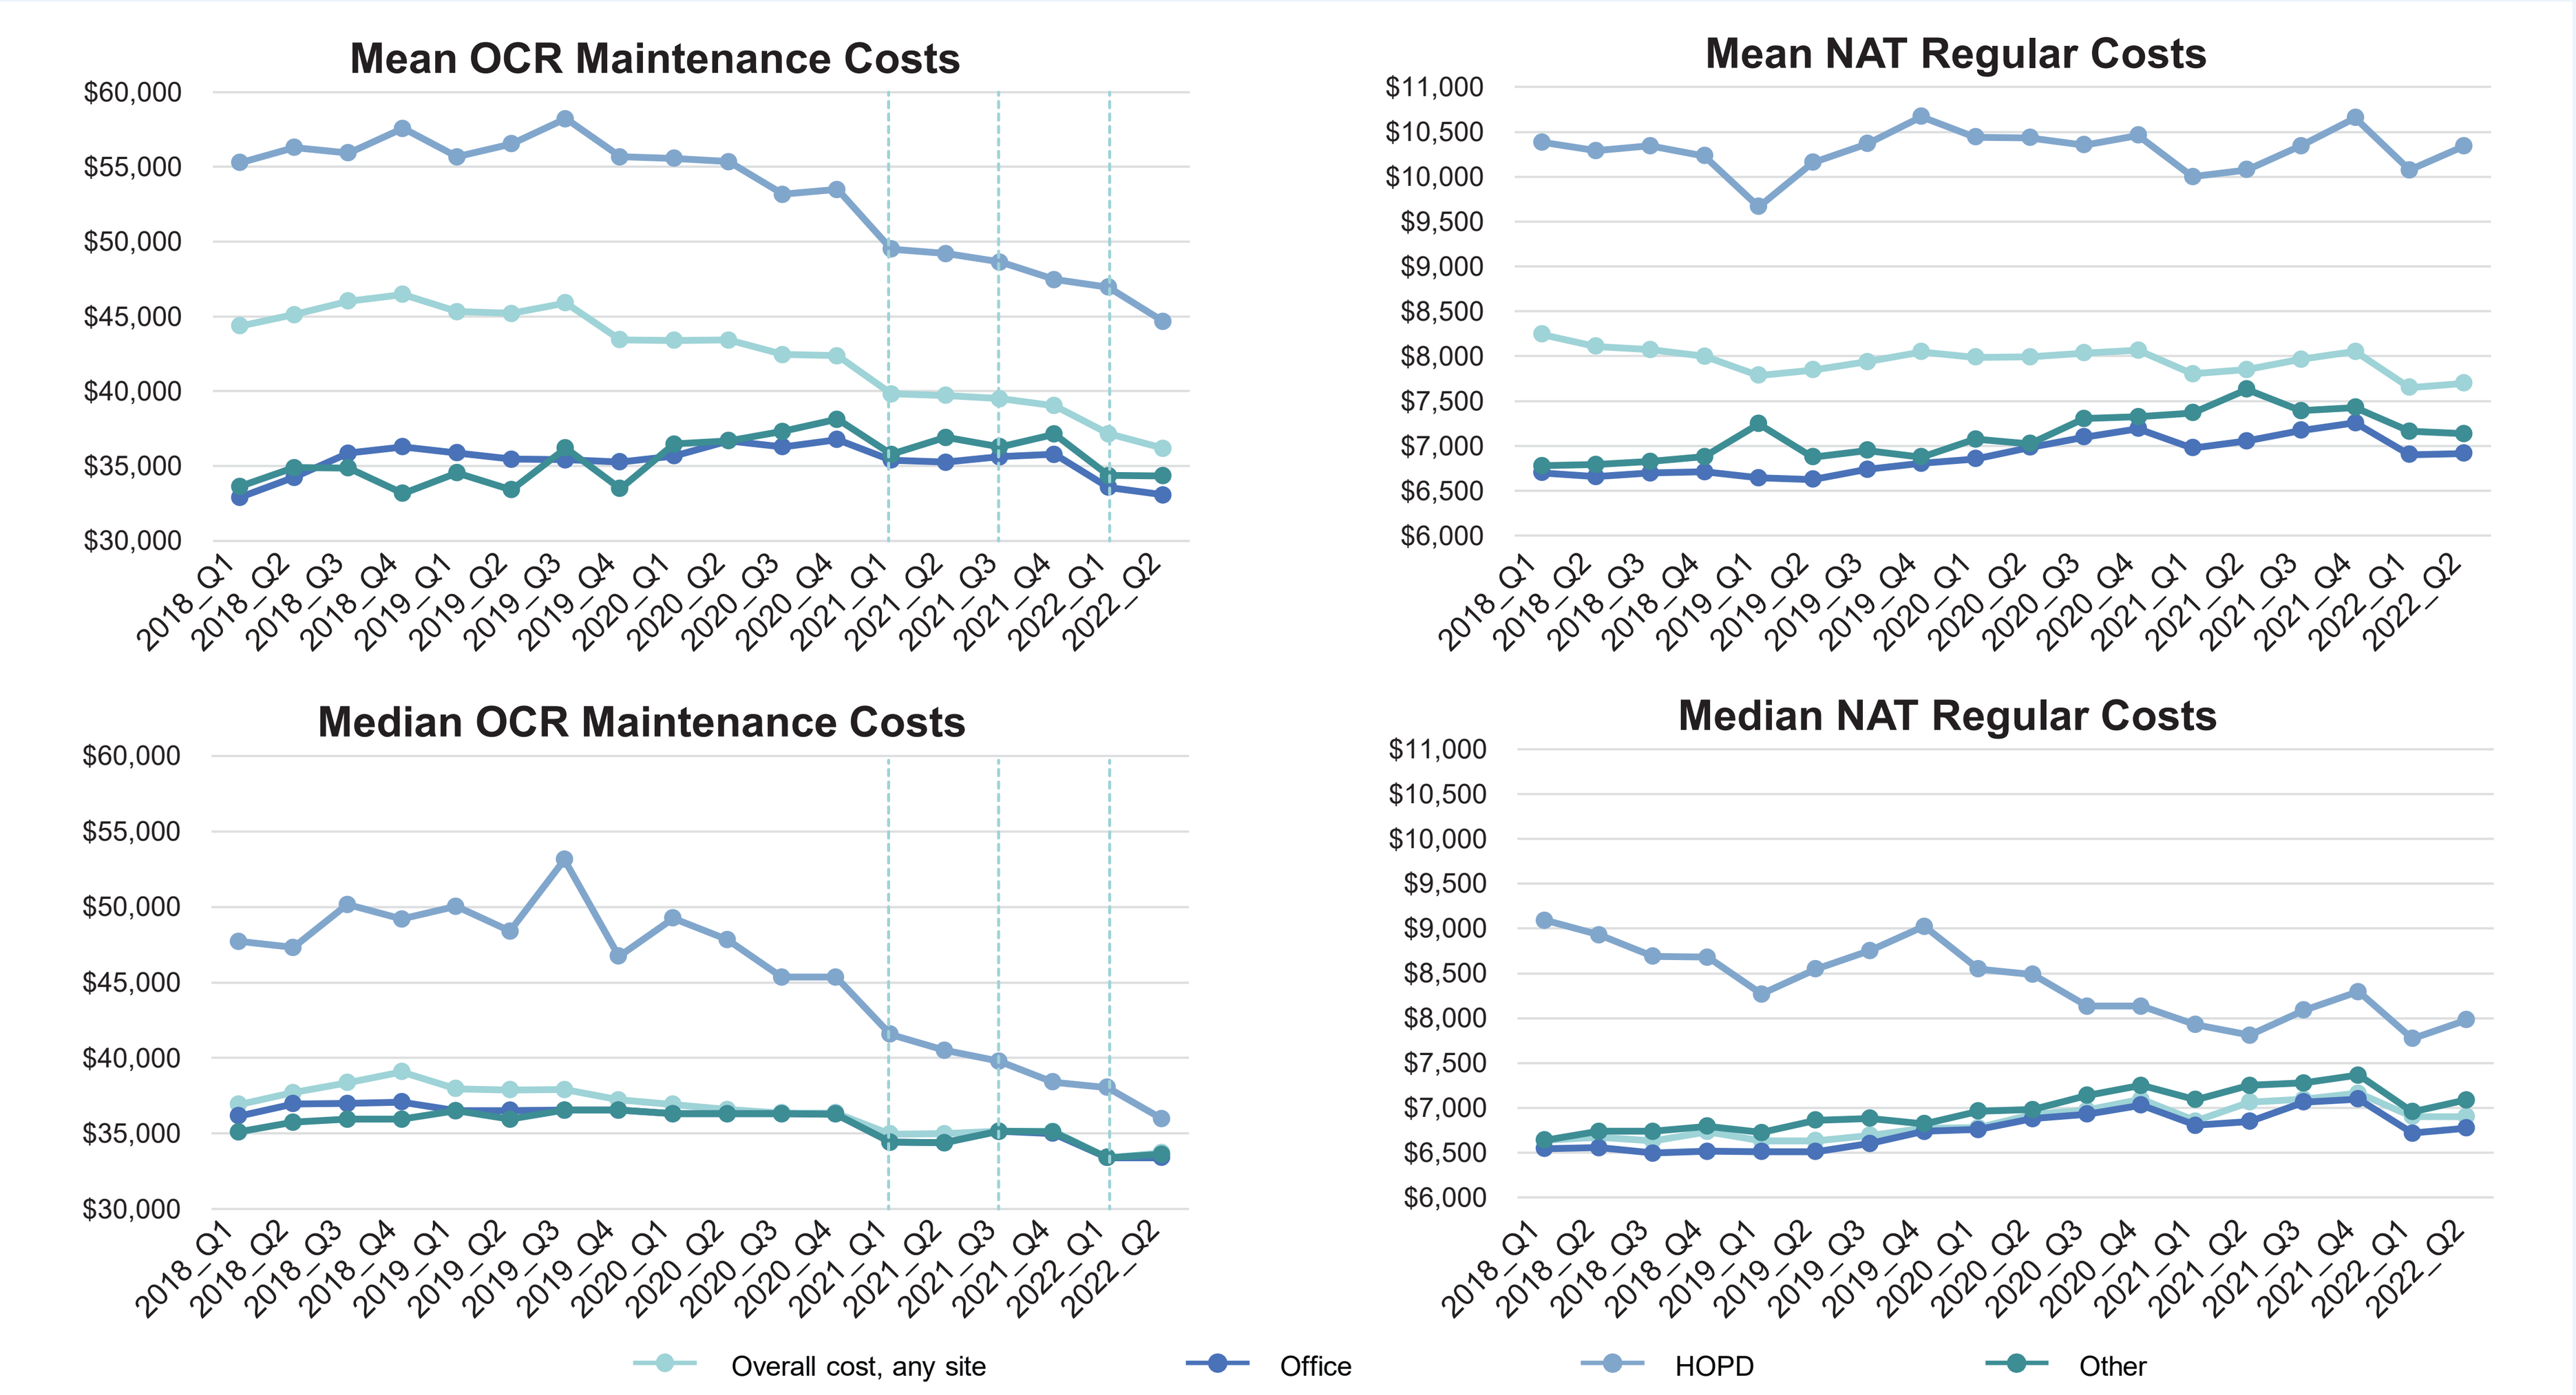
Mean OCR Maintenance Costs
Mean NAT Regular Costs
Median OCR Maintenance Costs
Median NAT Regular Costs
Overall cost, any site
Office
HOPD
Other

- Overall mean and median costs for OCR maintenance doses have decreased over time, primarily driven by a decrease in mean HOPD costs in addition to a shift in site of service from HOPD to office and other lower-cost settings
- There was a slight decrease in NAT overall mean cost while NAT HOPD mean costs remained relatively stable; costs at offices and other settings have slightly increased over time

HOPD, hospital outpatient department; NAT, natalizumab; OCR, ocrelizumab.
[a]Mean and median cost per dose adjusted to 2021 USD. Dotted lines indicate wholesale acquisition cost increases.

Figure 3. Percentage of OCR Maintenance Doses and NAT Regular Doses by Site of Service Over Time (2018 Q1–2022 Q2)

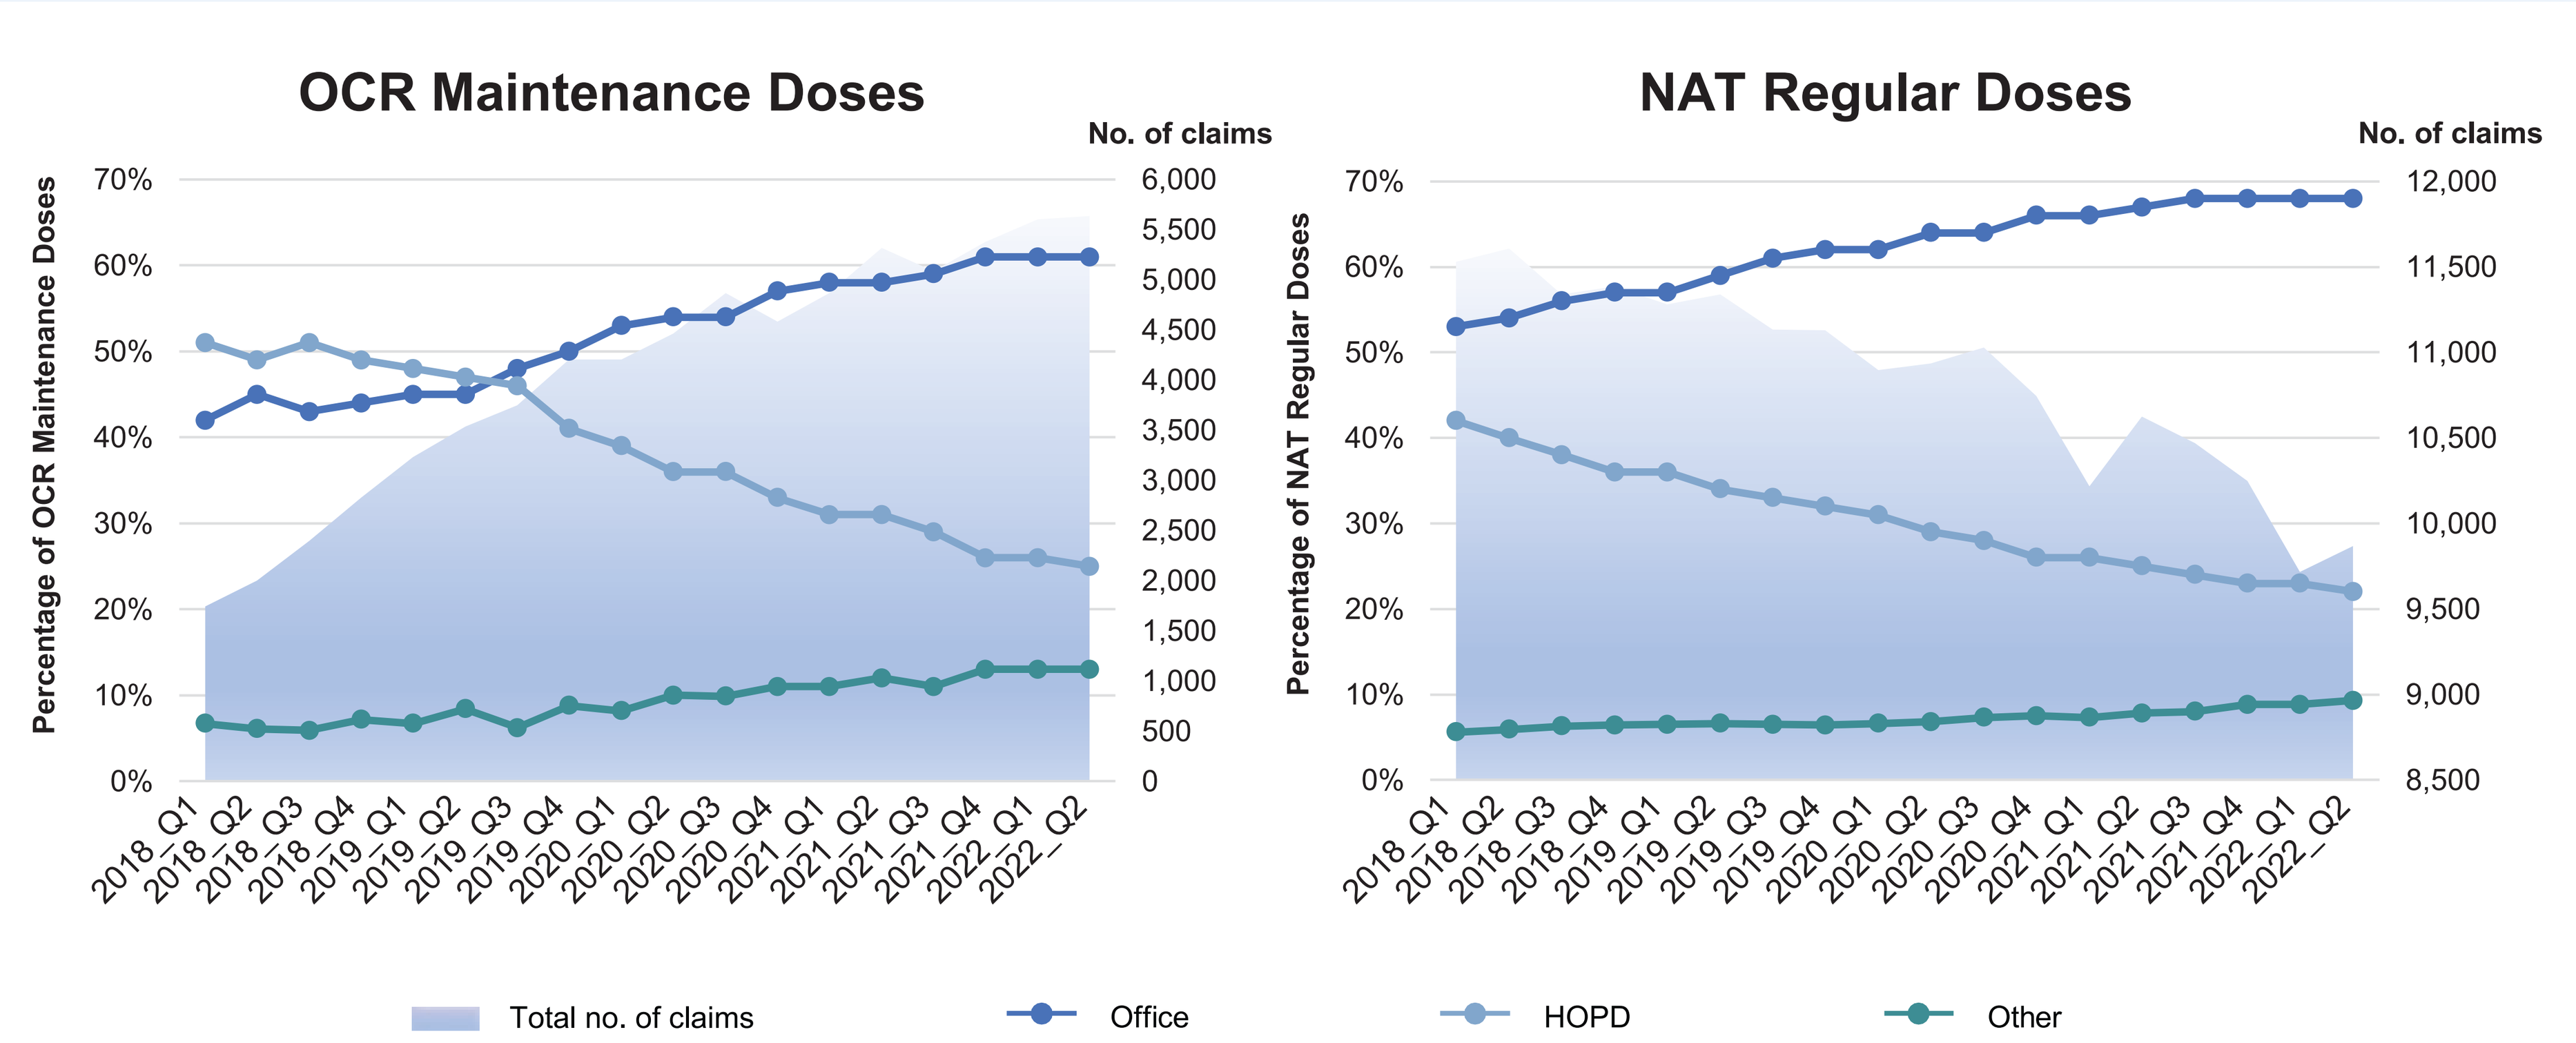
OCR Maintenance Doses
Percentage of OCR Maintenance Doses
No. of claims
NAT Regular Doses
Percentage of NAT Regular Doses
No. of claims
Total no. of claims
Office
HOPD
Other

- Proportions of OCR maintenance[a] and NAT regular doses infused at HOPD settings have decreased over time, while the proportions infused in office and other settings have increased over time

HOPD, hospital outpatient department; NAT, natalizumab; OCR, ocrelizumab.
[a]Proportions of OCR loading doses by site of service over time in **Supplement**.

Figure 5. Mean OOP Costs Per OCR Maintenance and NAT Regular Dose Over Time (2018–2021)[a,b]

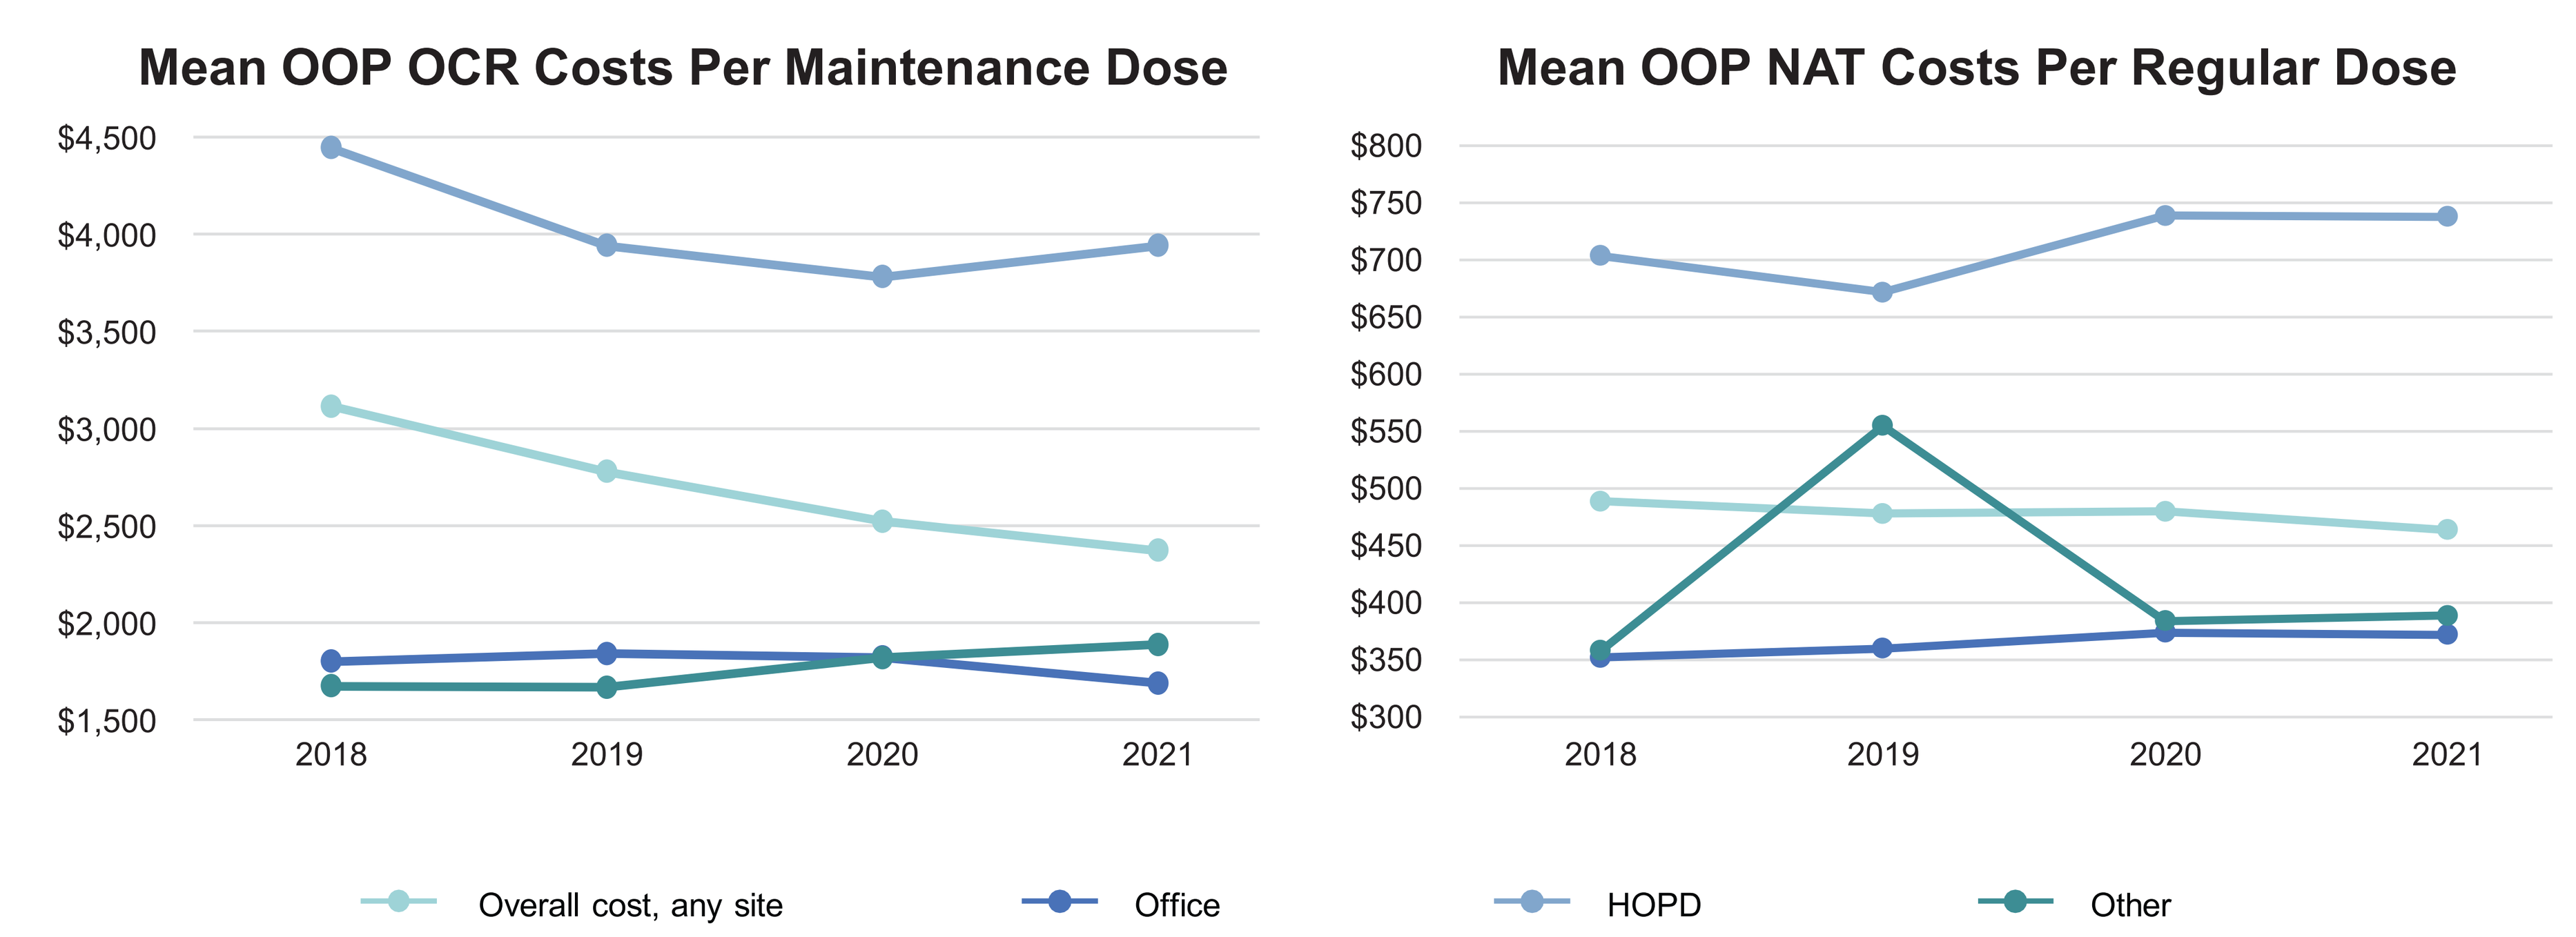
Mean OOP OCR Costs Per Maintenance Dose
Mean OOP NAT Costs Per Regular Dose
Overall cost, any site
Office
HOPD
Other

- From 2018, mean overall OOP costs of OCR per dose have been decreasing, whereas costs for NAT per dose have been relatively stable over time
- Mean OOP costs per dose are higher for OCR than NAT; however, the dosing schedule between OCR and NAT differs[c]

HOPD, hospital outpatient department; NAT, natalizumab; OCR, ocrelizumab; OOP, out-of-pocket.
[a]Mean cost per dose adjusted to 2021 USD.
[b]Data for 2022 not presented, because Q3-Q4 data for 2022 are unavailable as of April 2023.
[c]OCR maintenance doses administered every six months[2] and NAT regular doses administered every month.[4]

LIMITATIONS

- Reported costs are intended to be a representative national sample of commercially insured patients but may not generalize across all payers due to varying demographics, local practice patterns, negotiated reimbursement levels and other factors
- The PharMetrics Plus database provides a limited number of site-of-service categorizations. OCR is also infused in the home, stand-alone infusion centers and other settings, which the data do not provide, and limits our ability to present more granular descriptions for non-HOPD and non-office sites of service
- The analysis included patients who initiated OCR between January 1, 2018, and June 30, 2022, which included the time period immediately following the launch of OCR in the US. It is possible that the early users of OCR differ from patients who are currently treated with OCR, and these differences may influence the site of service at which patients receive treatment
- Additional limitations include potential for coding errors and missing data inherent to all claims analyses

CONCLUSIONS

- While site-of-service distribution for both OCR and NAT has shifted away from higher-cost HOPD to lower-cost office and other sites-of-service, reductions in mean OCR costs were higher than those for NAT over time
- Overall mean and median costs for OCR maintenance doses and NAT regular doses have decreased over time, primarily driven by a decrease in mean HOPD costs in addition to a shift in site of service from HOPD to office and other lower-cost settings
- From 2018, mean overall OOP costs of OCR by dose have been decreasing, whereas costs for NAT per dose have been increasing
- Real-world data can be leveraged to enable payers to make well-informed patient-access decisions for therapies while still supporting physician and patient choice

Presented at the 2023 Annual meeting of The Professional Society for Health Economics and Outcomes Research (ISPOR); May 7-10, 2023; Boston, MA, USA.

REFERENCES

1. Alvarez E, et al. *J Med Econ* 2023;27:1-30.
2. Ocrevus. Prescribing information. Genentech, Inc.; 2022.
3. Nicholas J, et al. *J Med Econ* 2020;23:885-93.
4. Tysabri. Prescribing information. Biogen, Inc.; 2021.
5. Perncezky J and Sellner J. *J Cent Nerv Syst Dis* 2022;14:11795735221135485.

DISCLOSURES

ED Pineda, A Patel, and C Ng are employees of Genentech, Inc. and shareholders of F. Hoffmann-La Roche Ltd.

This study was sponsored by F. Hoffmann-La Roche Ltd, Basel, Switzerland, editorial assistance was provided by Health Interactions, Inc., USA, and funded by F. Hoffmann-La Roche Ltd, Basel, Switzerland.

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission from CMSC or the authors of this poster.

https://bit.ly/3nSGeLz