#EE27

A novel cost impact analysis framework and model to evaluate medications for opioid use disorder within the US criminal justice system

Danmeng Huang[1] | Courtney Flynn[2] | Chris D Poole[1] | William Mullen[2] | Sabrina Gaiazov[2]
[1] Xcenda LLC (AmerisourceBergen), Carrollton, TX, USA | [2] Indivior, North Chesterfield, VA, USA

Objective

To develop a novel model framework to assess the cost impact of implementing a program to supply medications for opioid use disorder (MOUD) within the criminal justice EE27

system (CJS), accounting for the majority of likely costs but also plausible cost-offsets attributable to favorable outcomes.

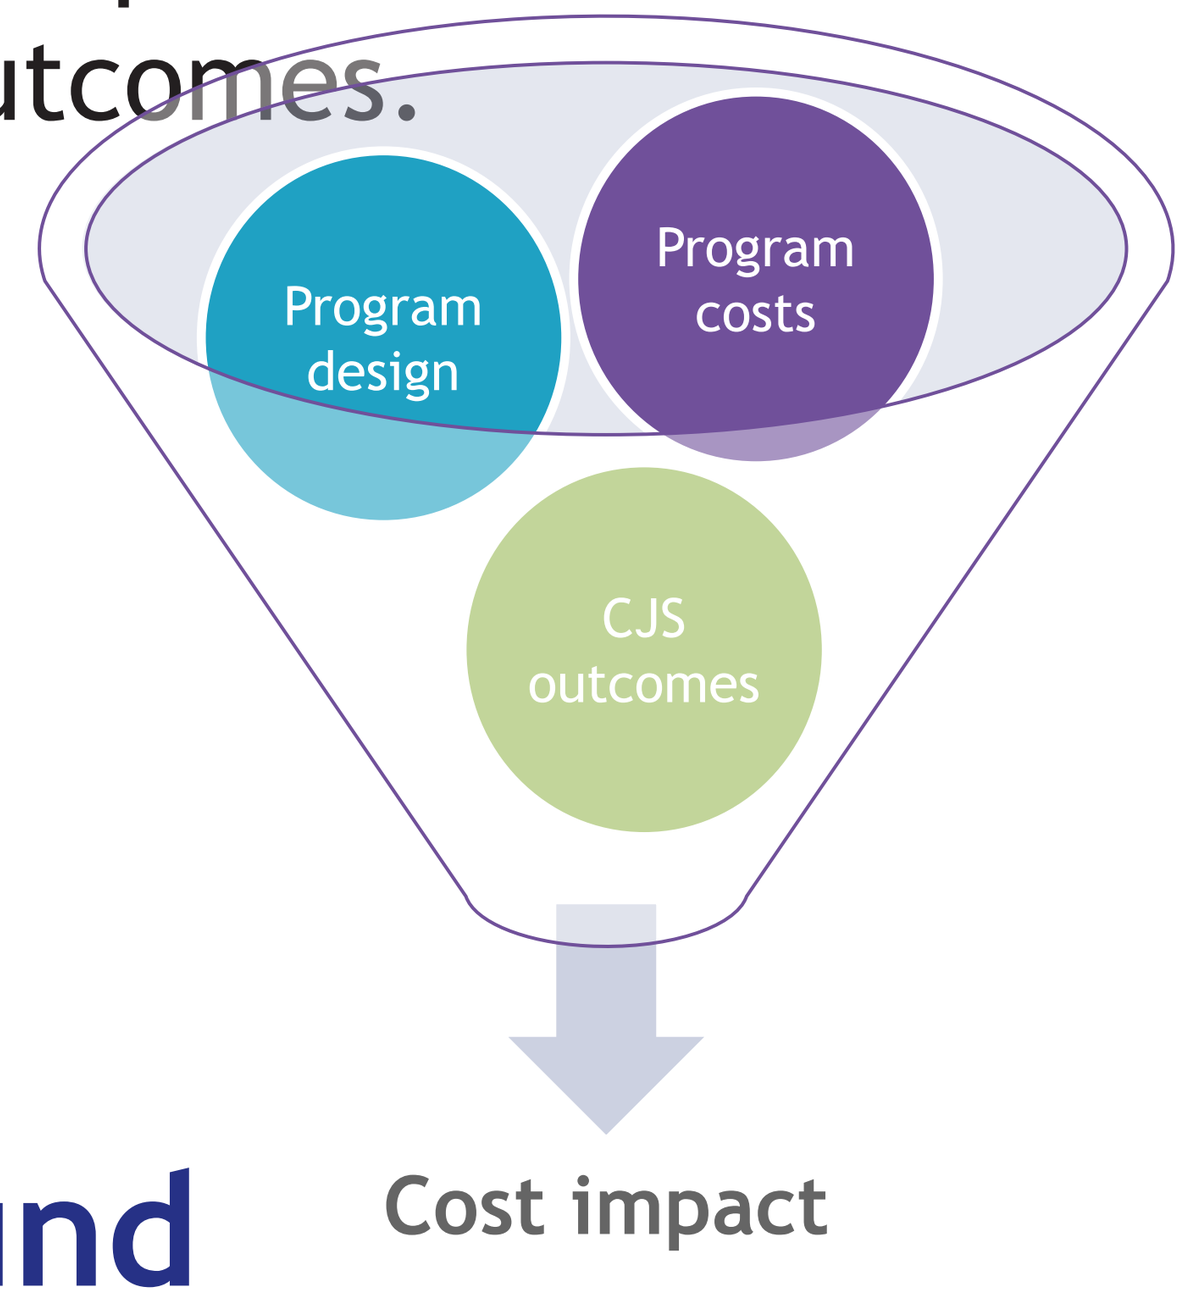
Program design
Program costs
CJS outcomes
Cost impact

Background

- Opioid use disorder (OUD) is common among inmates in United States jails and prisons, affecting around one-fifth of such persons[1].
- However, access to FDA-approved MOUDs is very limited; <1% of these institutions provide such care[2], despite the documented excess risks:
- People leaving prison without MOUD have a 40-fold higher risk of dying from overdose in the first 2 weeks following their discharge compared to the general population[3,4]
- MOUD in CJS has been shown to positively influence outcomes not only during, but also immediately following incarceration
- Despite these benefits, lack of funding is cited by CJS administrators as the principal barrier to establishing MOUD programs[5].
- Previous economic evaluations of MOUD in CJS have focused on acquisition costs and maintenance[6,7], while associated cost-offsets arising from improved outcomes are rarely estimated.

Methodology

- A literature review was conducted using Ovid MEDLINE, Emcare (via Ovid), Embase, Econlit, and EBM Reviews databases.
- Upon completion, 8 economic evaluations on MOUD programs within CJS were used.
- Correctional medical directors, clinical advisors and researchers were also engaged to provide input on model components, MOUD coverage and usability.
- Select inputs are modifiable to allow the user to tailor the model to their specific jail or prison.
- Outputs of the model include a summary of the cost impact of their selected MOUD program including total cost, per treated patient per month (PTPPM) and per incarcerated person per month (PPPM).
- Impact on drug diversion, violence, recidivism, and overdose over the selected time horizon are also included.

Model limitations

- The comparative clinical effectiveness of MOUD comparators are currently naïve since comprehensive bias-adjusted indirect treatment comparisons are unavailable in the CJS setting.
- Assumptions on the thresholds for step-fixed costs require validation by IDI's.
- The model framework is assumed to apply equally to the entirety of the incarcerated population and may not be sensitive to different facility types.
- The model framework is not yet equipped to extrapolate baseline characteristic drivers of CJS outcomes such as ethno-demography.

Model flow

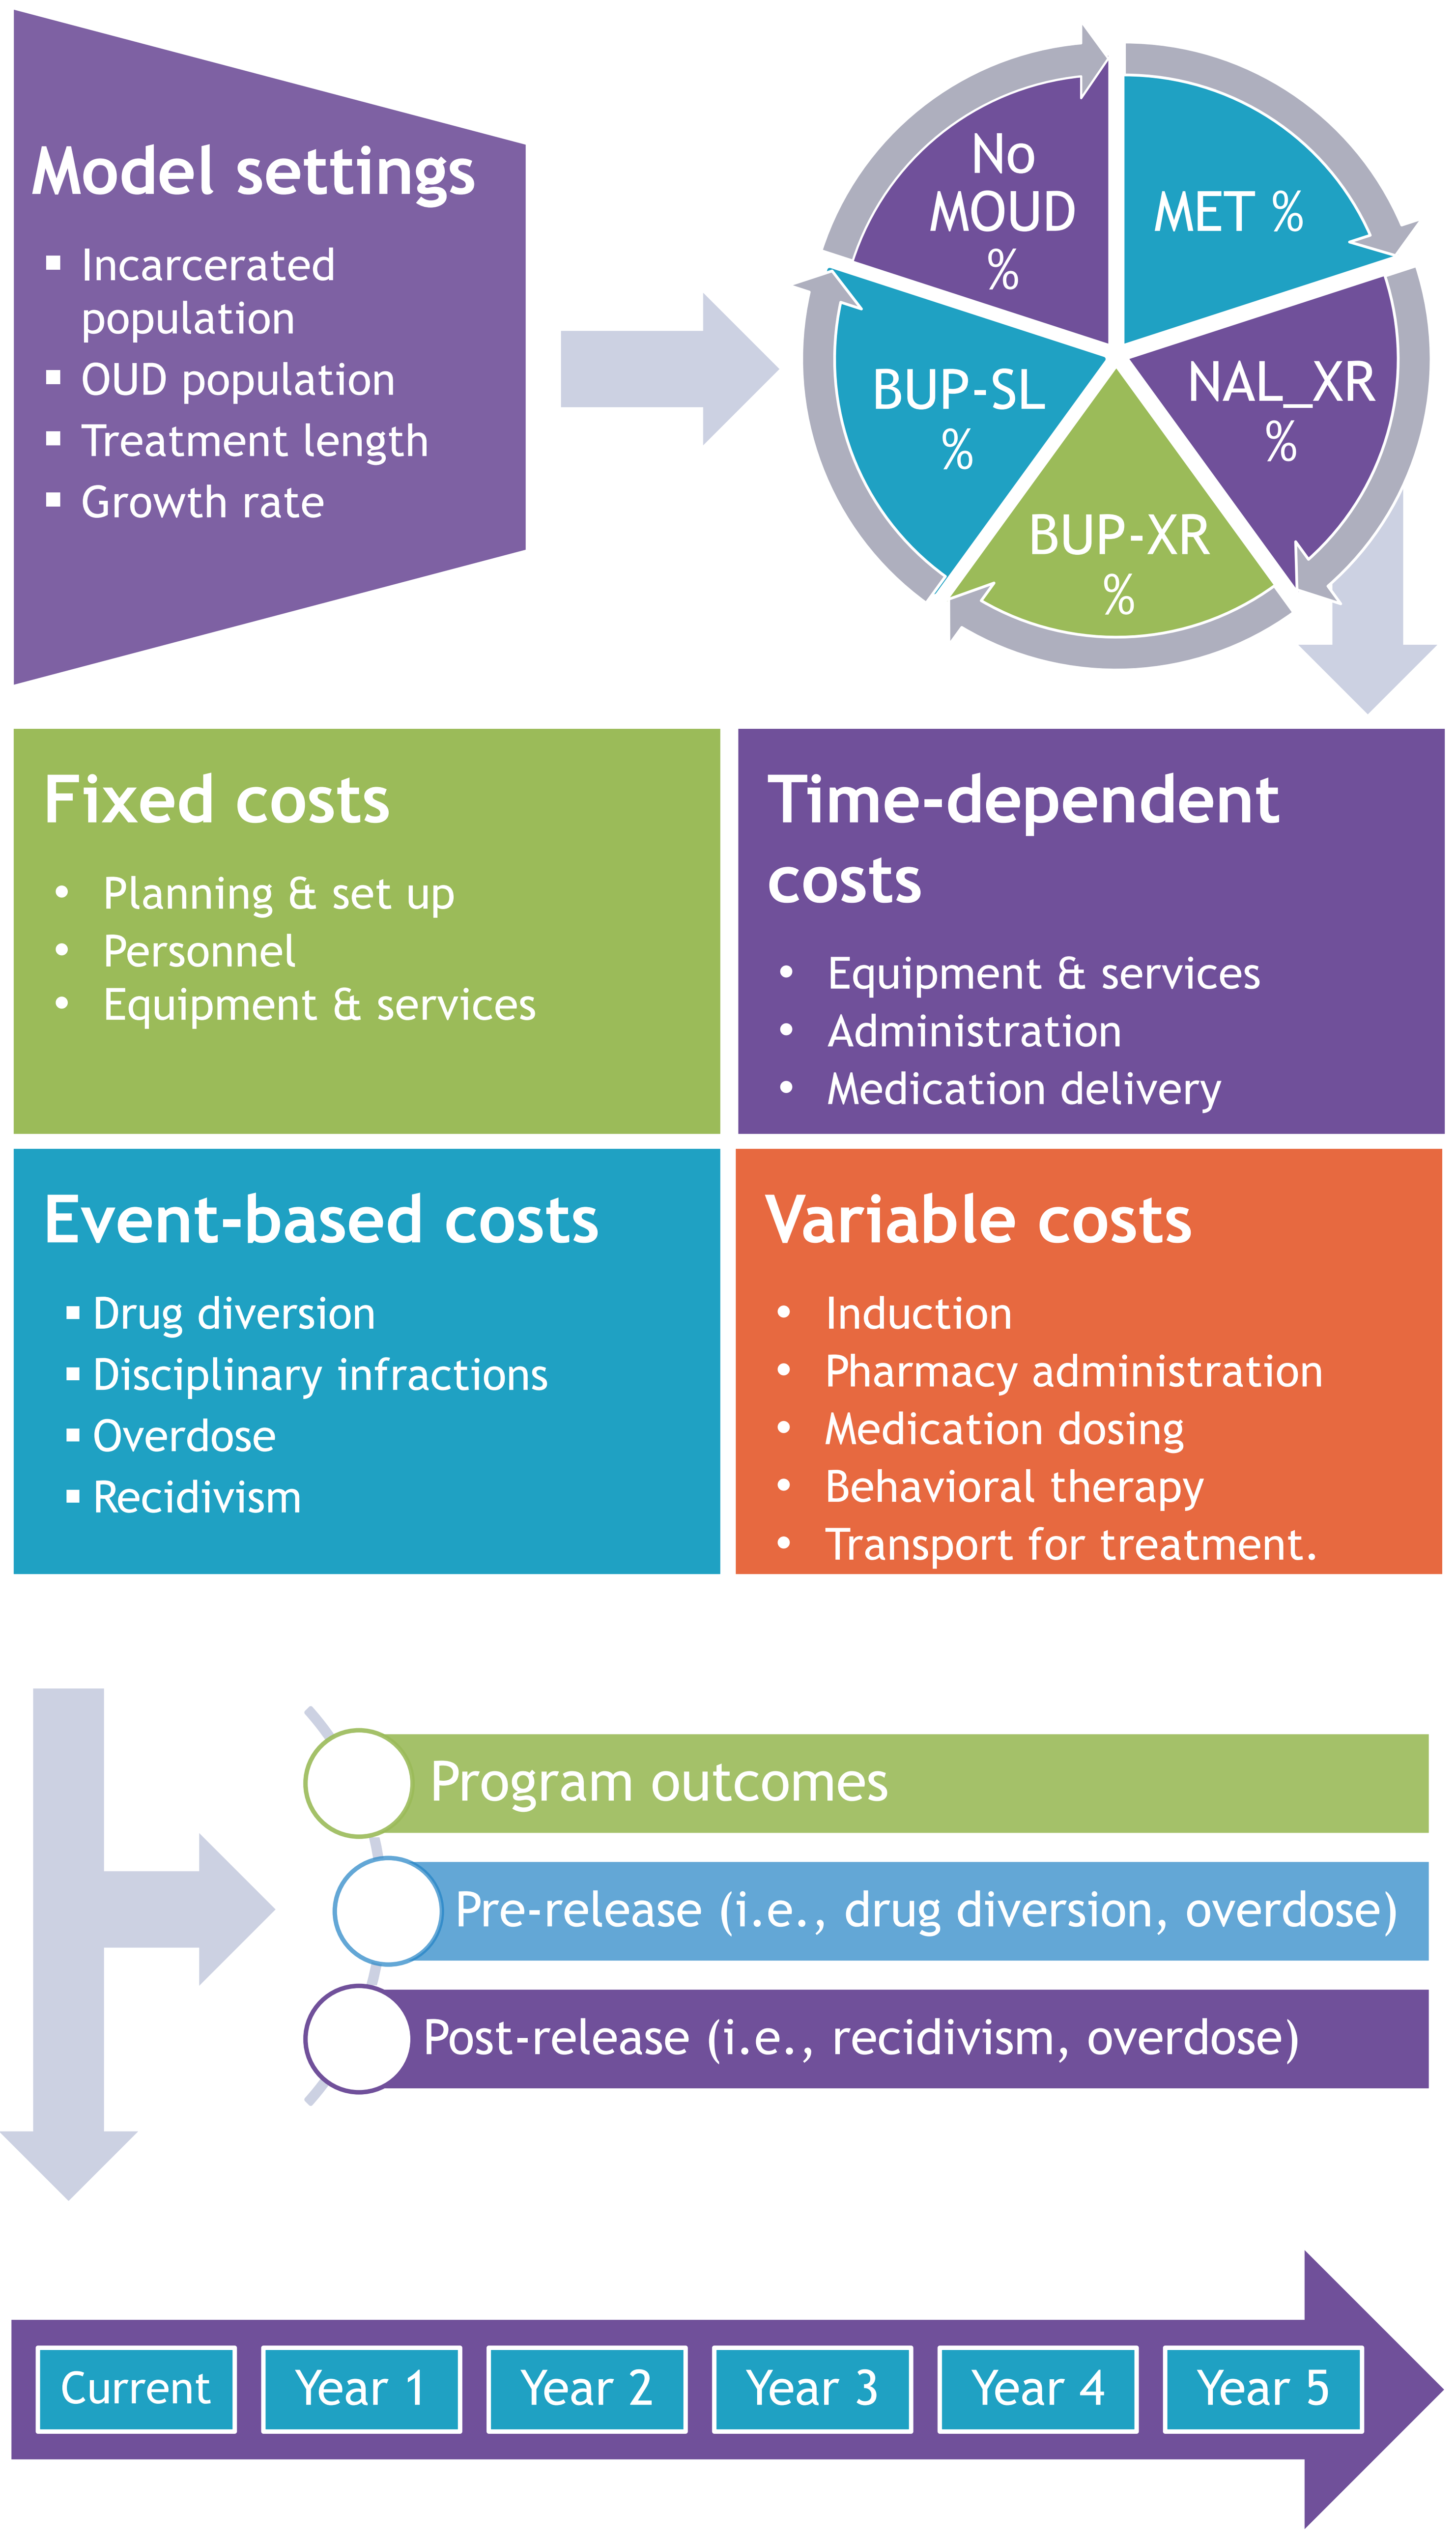
Model settings
Incarcerated population
OUD population
Treatment length
Growth rate
No MOUD %
MET %
NAL_XR %
BUP-XR %
BUP-SL %
Fixed costs
Planning & set up
Personnel
Equipment & services
Time-dependent costs
Equipment & services
Administration
Medication delivery
Event-based costs
Drug diversion
Disciplinary infractions
Overdose
Recidivism
Variable costs
Induction
Pharmacy administration
Medication dosing
Behavioral therapy
Transport for treatment.
Program outcomes
Pre-release (i.e., drug diversion, overdose)
Post-release (i.e., recidivism, overdose)
Current
Year 1
Year 2
Year 3
Year 4
Year 5

Future model applications

This cost impact analysis framework and model capture core components to comprehensively assess the economic impact of implementing different MOUD programs within CJS. Future research will consider a broader societal perspective including costs associated with lost productivity and premature mortality.

Disclosure

This study was designed and conducted by Xcenda, in collaboration with the sponsor, Indivior.

References

1. Bronson J, et al. Drug use, dependence, and abuse among state prisoners and jail inmates, 2007-2009 [Internet]. Available from: https://bjs.ojp.gov/content/pub/pdf/dudaspji0709.pdf **2.** Vestal C. New Momentum for Addiction Treatment Behind Bars [Internet]. The Pew Charitable Trusts; 2018. Available from: https://www.pewtrusts.org/en/research-and-analysis/blogs/stateline/2018/04/04/new-momentum-for-addiction-treatment-behind-bars **3.** Merrall ELC, et al. Meta-analysis of drug-related deaths soon after release from prison. Addiction. 2010 Sep;105(9):1545–54. **4.** Ranapurwala SI, et al. Opioid Overdose Mortality Among Former North Carolina Inmates: 2000-2015. Am J Public Health. 2018 Sep;108(9):1207–13. **5.** Scott CK, et al. Availability of best practices for opioid use disorder in jails and related training and resource needs: findings from a national interview study of jails in heavily impacted counties in the U.S. Health Justice. 2022 Dec 20;10(1):36. **6.** Ryan DA, et al. Budget impact tool for the incorporation of medications for opioid use disorder into jail/prison facilities. J Subst Use Addict Treat. 2023 Mar;146:208943. **7.** Medication-Assisted Treatment (MAT) for Opioid Use Disorder in Jails and Prisons: A Planning and Implementation Toolkit [Internet]. National Council for Mental Wellbeing. 2022. Available from: https://www.thenationalcouncil.org/resources/medication-assisted-treatment-mat-for-opioid-use-disorder-in-jails-and-prisons-a-planning-and-implementation-toolkit/

Presented at: International Society for Pharmacoeconomics and Outcomes Research Annual Meeting (ISPOR 2023)
May 7-10, 2023 | Boston, MA, USA