Association of Clinical Response Criteria and Disease Activity Levels with Physical Function and HRQoL in Patients with Axial Spondyloarthritis: 52-Week Results from Two Phase 3 Randomized, Placebo-Controlled Studies

Marina Magrey,[1] Atul Deodhar,[2] Philip J. Mease,[3] Victoria Navarro-Compán,[4] Sofia Ramiro,[5] Martin Rudwaleit,[6] Christine de la Loge,[7] Carmen Fleurinck,[8] Vanessa Taieb,[9] Michael Frank Mørup,[10] Ute Massow,[11] Jonathan Kay[12]

Presented at ISPOR 2023 | May 7–10 | Boston, MA, USA

PCR68

Objectives

To examine how reaching different clinical response criteria and disease activity levels translates into physical function and health-related quality of life (HRQoL) improvements in patients with axial spondyloarthritis (axSpA).

Background

- Bimekizumab (BKZ) is a monoclonal IgG1 antibody that selectively inhibits interleukin (IL)-17F in addition to IL-17A.
- BKZ has demonstrated sustained efficacy and was well tolerated up to 52 weeks in the phase 3 studies BE MOBILE 1 (NCT03928704; non-radiographic [nr]-axSpA) and BE MOBILE 2 (NCT03928743; radiographic [r]-axSpA, i.e., ankylosing spondylitis).[1,2]
- Here, we present a post hoc analysis of Week 52 results from these phase 3 studies, covering the full spectrum of axSpA independent of treatment arm.

Methods

- All patients reaching the following criteria at Week 52 were pooled regardless of initial treatment arm (placebo/BKZ 160mg Q4W), by study:
 - **ASAS response:** ASAS20 not reached (<ASAS20), ASAS20 reached but not ASAS40 (ASAS20–<ASAS40), ASAS40 reached (ASAS40)
 - **ASAS partial remission (ASAS PR):** no, yes
 - **Disease activity levels (ASDAS):** very high disease activity/high disease activity (vHDA/HDA; ≥2.1), low disease activity (LDA; ≥1.3–<2.1), inactive disease (ID; <1.3)
 - **ASDAS change from baseline (CfB):** no response (ASDAS CfB <1.1), clinically important improvement (ASDAS-CII; ASDAS CfB <2.0–≥1.1), major improvement (ASDAS-MI; ASDAS CfB ≥2.0)
- Associations between achievement of these criteria and changes in the following patient-reported outcome measures were assessed:
 - Physical function: **BASFI** (a component of ASAS response criteria), **SF-36 PCS**
 - HRQoL: **ASQoL, EQ-5D-3L utilities** (UK tariff)
- Observed case data are reported.

Results

Patients

- Of randomized patients, 220/254 (87%) with nr-axSpA and 298/332 (90%) with r-axSpA completed Week 52.
- Baseline characteristics were generally similar between patients with nr-axSpA and r-axSpA, except for sex, symptom duration, time since first diagnosis, HLA-B27 positivity, hs-CRP, and prior tumor necrosis factor inhibitor exposure (**Table 1**).
- Nevertheless, baseline scores in physical function and HRQoL measures indicated similar burden of clinical symptoms between the two patient populations (**Table 1**).

Association of Clinical Response and Disease Activity with Physical Function and HRQoL

- Patients achieving higher ASAS response levels demonstrated sequentially greater mean improvements from baseline in BASFI (**Figure 1A**) and SF-36 PCS (**Figure 1B**) scores across the axSpA disease spectrum.
- Achievement of higher ASAS response levels was also associated with greater improvements in ASQoL and EQ-5D-3L (**Figure 2**).
- Similar associations were observed for ASAS PR, ASDAS disease activity states, and levels of ASDAS improvement (**Figures 1–2**).
- In all cases, improvements in BASFI, SF-36 PCS, ASQoL, and EQ-5D-3L were of similar amplitude between patients with nr-axSpA and r-axSpA (**Figures 1–2**).

Conclusions

Patients with axSpA treated with bimekizumab who achieved increasingly stringent clinical response criteria and lower disease activity levels at Week 52 reported greater improvements in physical function and HRQoL. Similar amplitudes of improvement were observed across the axSpA disease spectrum.

Summary

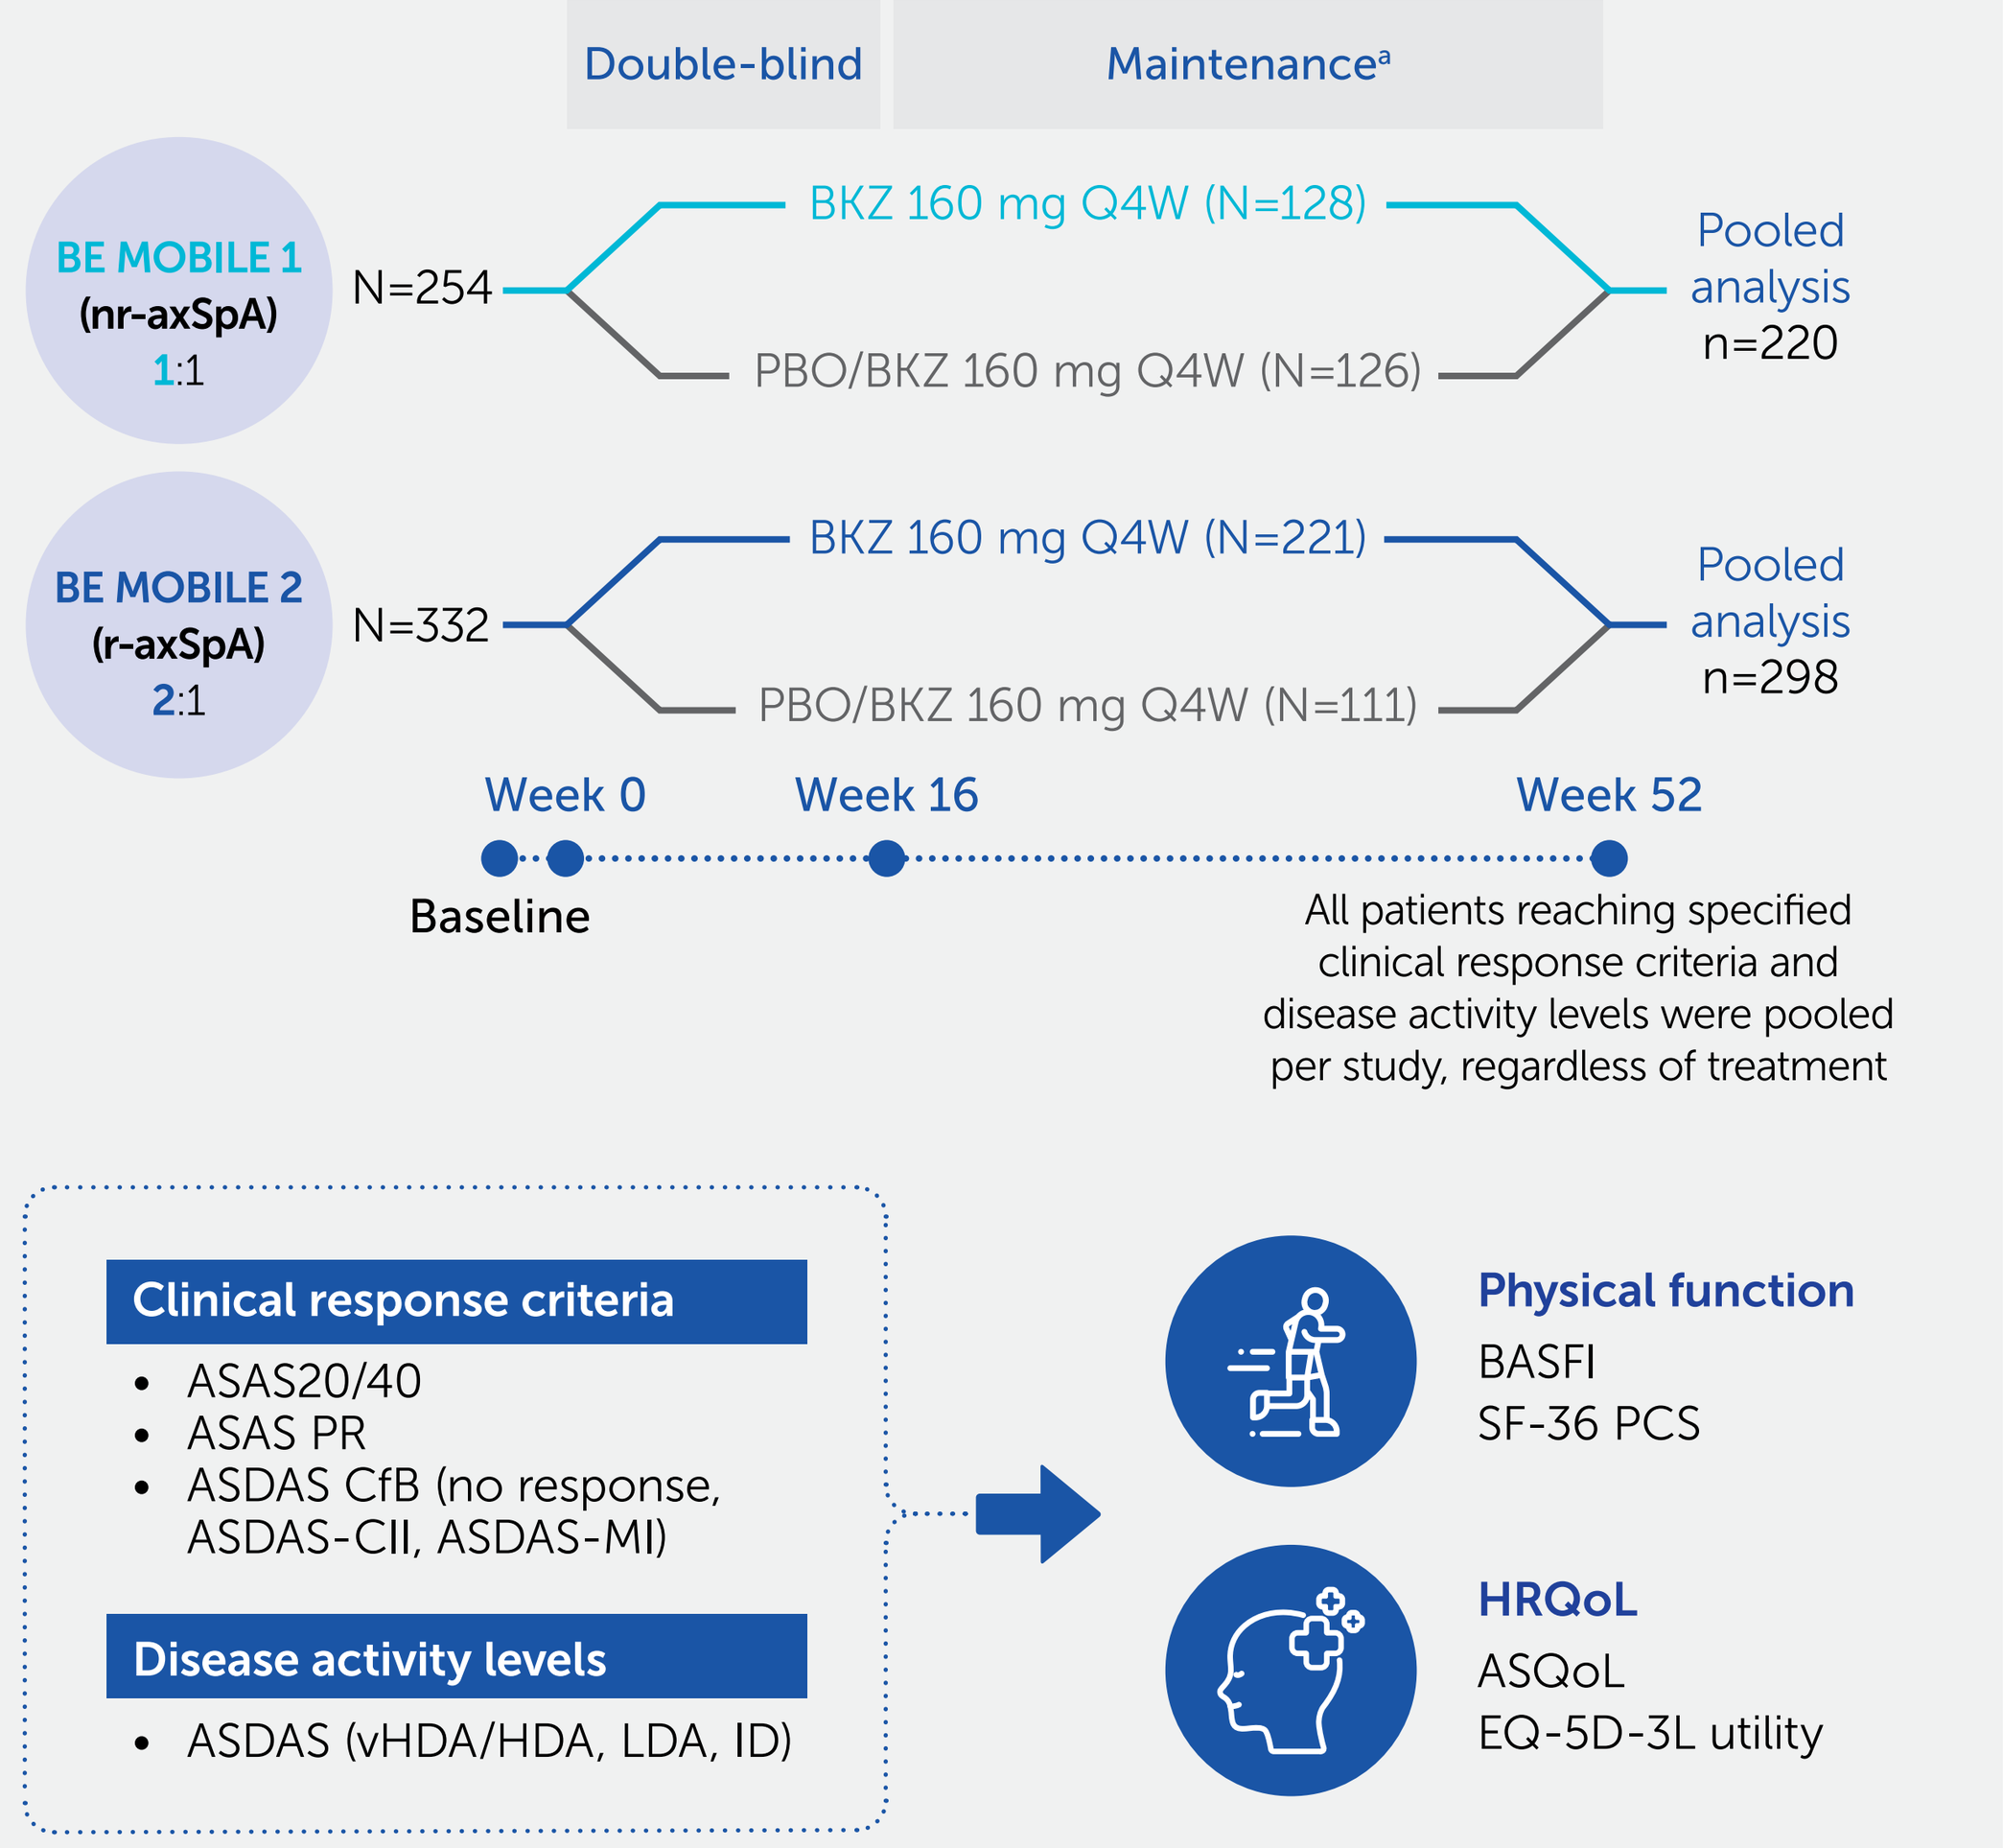
Double-blind
Maintenance[a]
BE MOBILE 1 (nr-axSpA) 1:1
N=254
BKZ 160 mg Q4W (N=128)
PBO/BKZ 160 mg Q4W (N=126)
Pooled analysis n=220
BE MOBILE 2 (r-axSpA) 2:1
N=332
BKZ 160 mg Q4W (N=221)
PBO/BKZ 160 mg Q4W (N=111)
Pooled analysis n=298
Week 0
Week 16
Week 52
Baseline
All patients reaching specified clinical response criteria and disease activity levels were pooled per study, regardless of treatment
Clinical response criteria
ASAS20/40
ASAS PR
ASDAS CfB (no response, ASDAS-CII, ASDAS-MI)
Disease activity levels
ASDAS (vHDA/HDA, LDA, ID)
Physical function
BASFI
SF-36 PCS
HRQoL
ASQoL
EQ-5D-3L utility

Patients with active axSpA who achieved increasingly stringent clinical response criteria and lower levels of disease activity at Week 52 reported greater improvements in physical function and HRQoL

[a]From Week 16, all patients were treated with BKZ 160mg Q4W, irrespective of baseline treatment randomization.

Table 1 Patient demographics and baseline characteristics

	BE MOBILE 1 (nr-axSpA) N=254	BE MOBILE 2 (r-axSpA) N=332
Age, years	39.4 (11.5)	40.4 (12.3)
Sex, male, n (%)	138 (54.3)	240 (72.3)
HLA-B27 positive, n (%)	197 (77.6)	284 (85.5)
Symptom duration, years	9.0 (8.8)	13.5 (10.3)
Time since first diagnosis of axSpA, years	3.6 (5.8)	6.4 (7.9)
ASDAS	3.7 (0.7)	3.7 (0.8)[a]
BASDAI	6.8 (1.3)	6.5 (1.3)
hs-CRP, mg/L, median (min, max)	6.3 (0.1, 79.1)	7.4 (0.1, 105.4)
hs-CRP >ULN,[b,c] n (%)	141 (55.5)	204 (61.4)
Total spinal pain score	7.2 (1.5)	7.2 (1.5)
BASFI	5.4 (2.3)	5.2 (2.1)
SF-36 PCS	33.4 (8.5)	34.4 (8.5)
ASQoL	9.4 (4.5)	8.9 (4.6)
EQ-5D-3L utility score,[d]	0.53 (0.28)	0.54 (0.29)
Prior TNFi exposure, n (%)	27 (10.6)	54 (16.3)

Randomized set. All variables reported as mean (SD) unless specified otherwise. [a]n=331; [b]ULN value for hs-CRP was 5 mg/L; [c]In BE MOBILE 1, patients had elevated hs-CRP ≥6.0 mg/L; [d]Utility scores were derived using the UK value set.

Figure 1 Achievement of increasingly stringent clinical response criteria and lower disease activity levels is associated with greater improvements in physical function

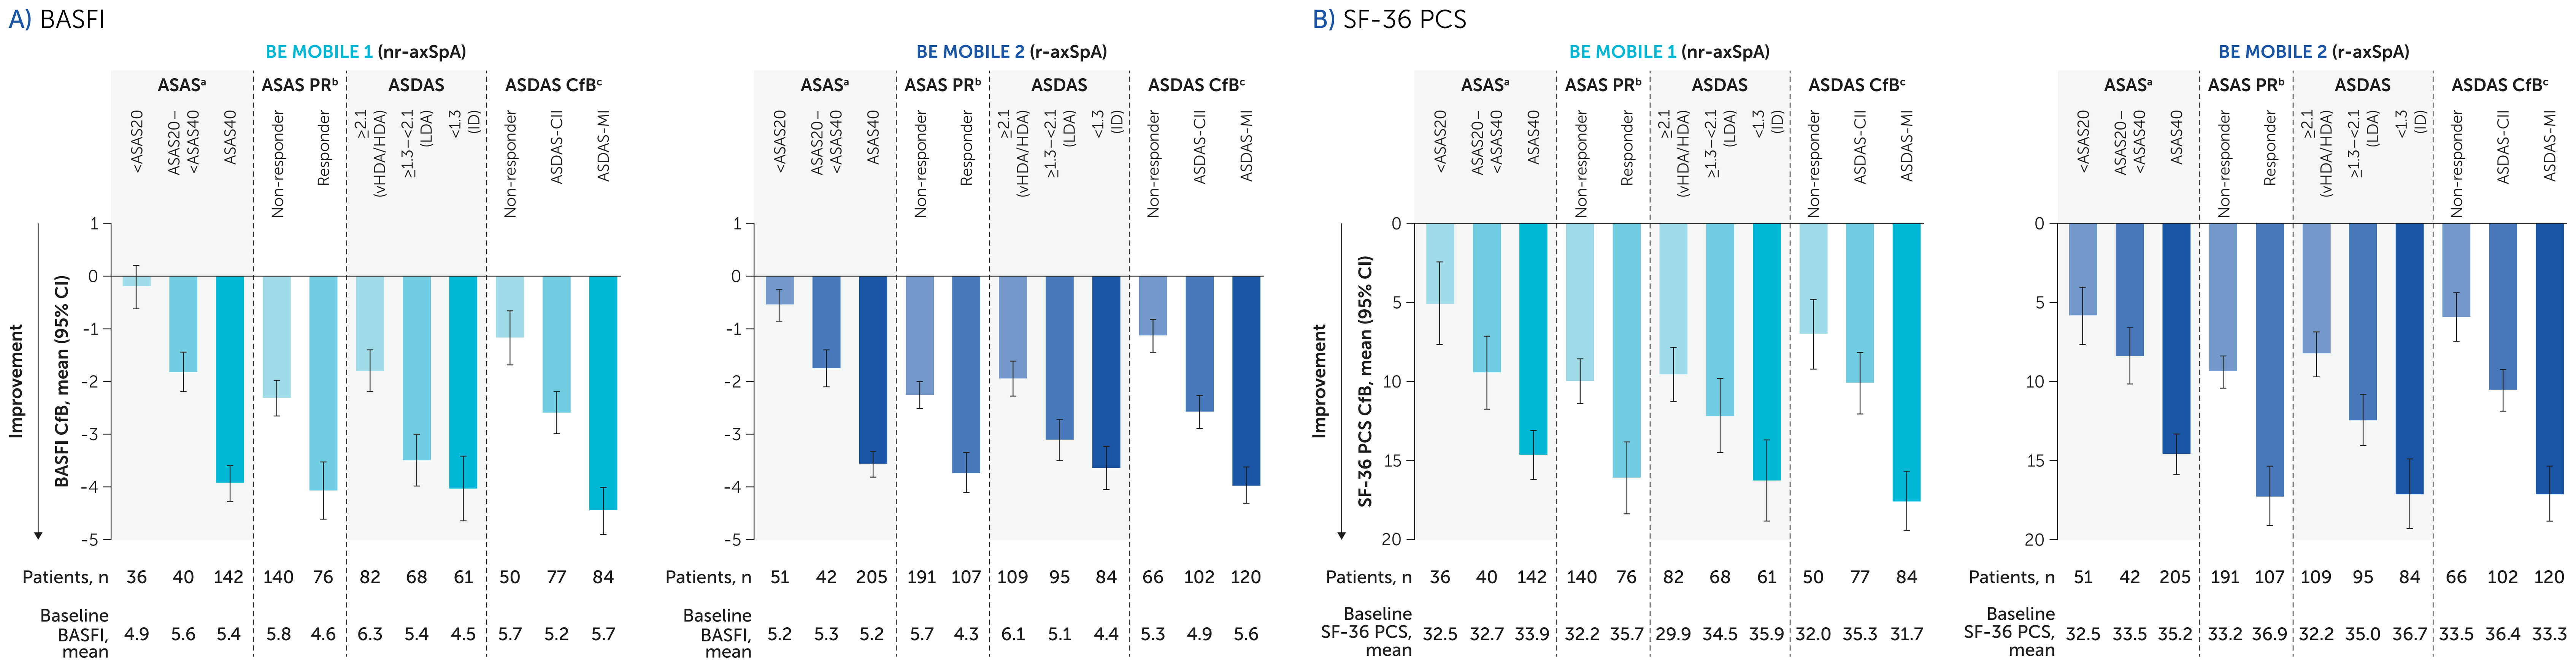
A) BASFI
B) SF-36 PCS
BE MOBILE 1 (nr-axSpA)
BE MOBILE 2 (r-axSpA)
ASAS[a]
ASAS PR[b]
ASDAS
ASDAS CfB[c]
<ASAS20
ASAS20–<ASAS40
ASAS40
Non-responder
Responder
≥2.1 (vHDA/HDA)
≥1.3–<2.1 (LDA)
<1.3 (ID)
Non-responder
ASDAS-CII
ASDAS-MI
Improvement
BASFI CfB, mean (95% CI)
SF-36 PCS CfB, mean (95% CI)
Patients, n 36 40 142 140 76 82 68 61 50 77 84
Baseline BASFI, mean 4.9 5.6 5.4 5.8 4.6 6.3 5.4 4.5 5.7 5.2 5.7
Patients, n 51 42 205 191 107 109 95 84 66 102 120
Baseline BASFI, mean 5.2 5.3 5.2 5.7 4.3 6.1 5.1 4.4 5.3 4.9 5.6
Patients, n 36 40 142 140 76 82 68 61 50 77 84
Baseline SF-36 PCS, mean 32.5 32.7 33.9 32.2 35.7 29.9 34.5 35.9 32.0 35.3 31.7
Patients, n 51 42 205 191 107 109 95 84 66 102 120
Baseline SF-36 PCS, mean 32.5 33.5 35.2 33.2 36.9 32.2 35.0 36.7 33.5 36.4 33.3

Figure 2 Achievement of increasingly stringent clinical response criteria and lower disease activity levels is associated with greater improvements in HRQoL

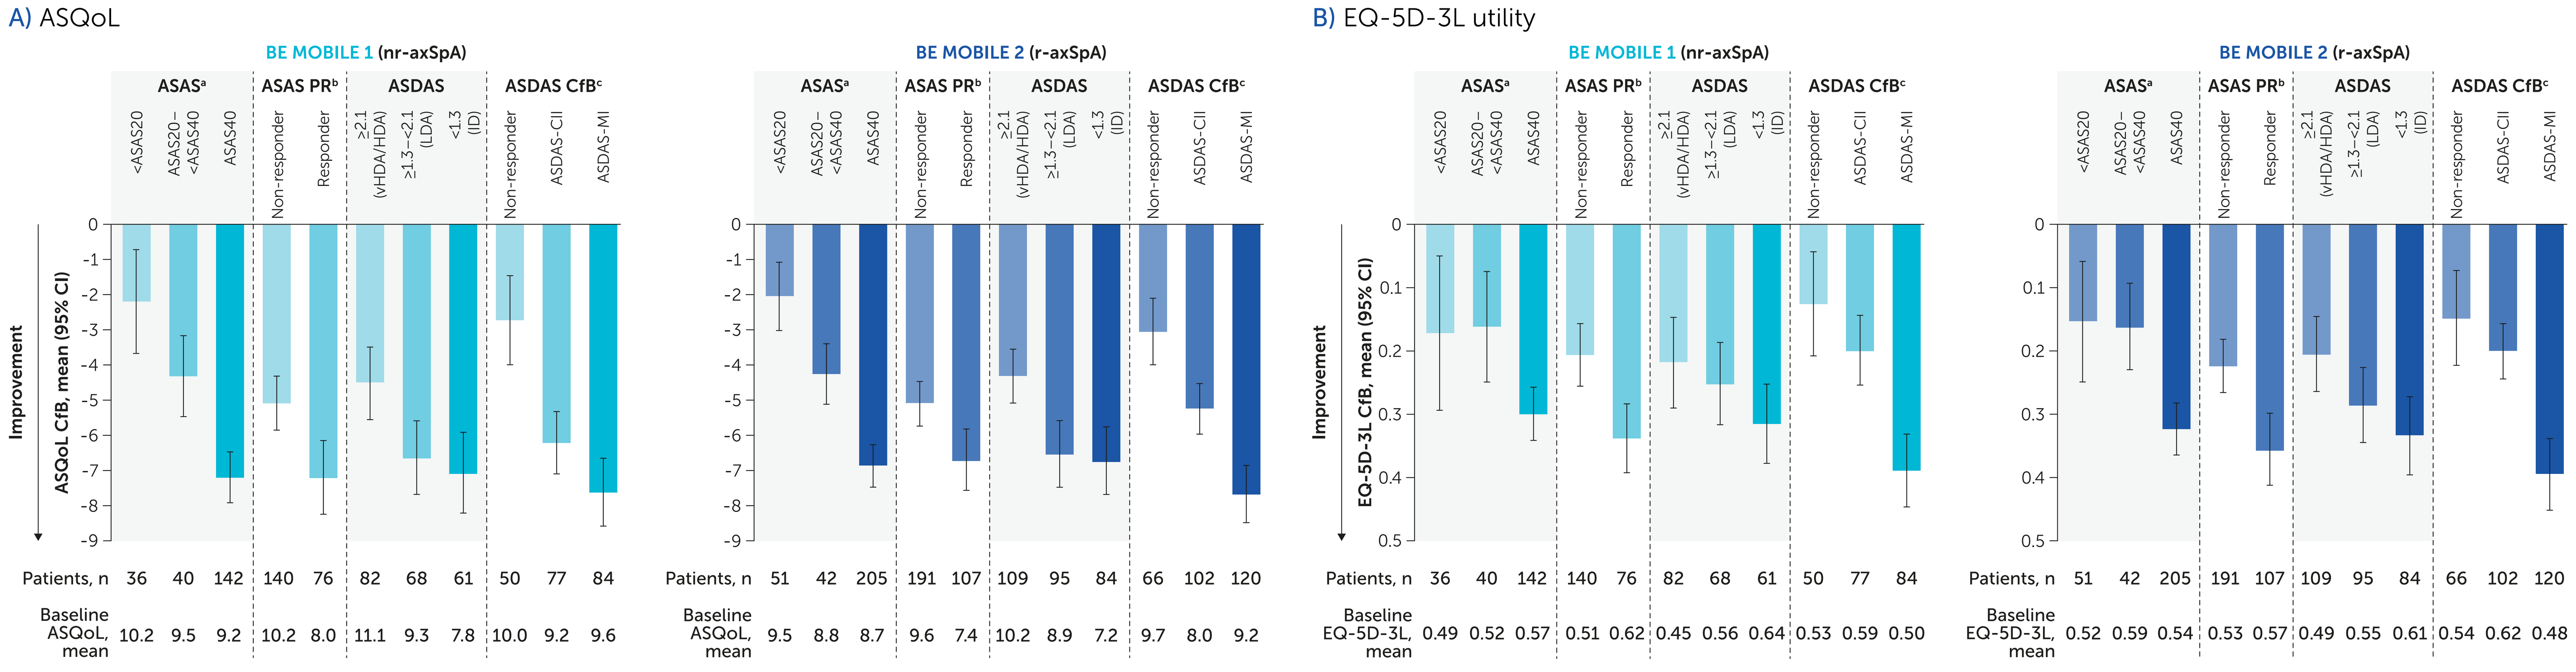
A) ASQoL
B) EQ-5D-3L utility
BE MOBILE 1 (nr-axSpA)
BE MOBILE 2 (r-axSpA)
ASAS[a]
ASAS PR[b]
ASDAS
ASDAS CfB[c]
<ASAS20
ASAS20–<ASAS40
ASAS40
Non-responder
Responder
≥2.1 (vHDA/HDA)
≥1.3–<2.1 (LDA)
<1.3 (ID)
Non-responder
ASDAS-CII
ASDAS-MI
Improvement
ASQoL CfB, mean (95% CI)
EQ-5D-3L CfB, mean (95% CI)
Patients, n 36 40 142 140 76 82 68 61 50 77 84
Baseline ASQoL, mean 10.2 9.5 9.2 10.2 8.0 11.1 9.3 7.8 10.0 9.2 9.6
Patients, n 51 42 205 191 107 109 95 84 66 102 120
Baseline ASQoL, mean 9.5 8.8 8.7 9.6 7.4 10.2 8.9 7.2 9.7 8.0 9.2
Patients, n 36 40 142 140 76 82 68 61 50 77 84
Baseline EQ-5D-3L, mean 0.49 0.52 0.57 0.51 0.62 0.45 0.56 0.64 0.53 0.59 0.50
Patients, n 51 42 205 191 107 109 95 84 66 102 120
Baseline EQ-5D-3L, mean 0.52 0.59 0.54 0.53 0.57 0.49 0.55 0.61 0.54 0.62 0.48

Data reported as observed case. Categories are mutually exclusive. [a]Patients were stratified into three mutually exclusive groups based on ASAS clinical response criteria: did not achieve ASAS20 (<ASAS20), achieved ASAS20 (≥20% reduction and absolute reduction of ≥1 unit in ≥3 of four ASAS domains, and no deterioration in the remaining domain) but did not achieve ASAS40 (≥40% reduction and absolute reduction of ≥2 units in ≥3 of the four ASAS domains and no deterioration in the remaining domain; ASAS20–<ASAS40), and achieved ASAS40; [b]ASAS-PR: achievement of a score ≤2 units across all four ASAS domains; [c]Patients were stratified into three mutually exclusive groups based on ASDAS CfB.

ASAS: Assessment of SpondyloArthritis International Society; **ASAS PR:** ASAS partial remission; **ASDAS:** Ankylosing Spondylitis Disease Activity Score; **ASDAS-CII:** ASDAS clinically important improvement; **ASDAS-MI:** ASDAS major improvement; **ASQoL:** Ankylosing Spondylitis Quality of Life; **axSpA:** axial spondyloarthritis; **BASDAI:** Bath Ankylosing Spondylitis Disease Activity Index; **BASFI:** Bath Ankylosing Spondylitis Functional Index; **BKZ:** bimekizumab; **CfB:** change from baseline; **CI:** confidence interval; **CRP:** C-reactive protein; **EQ-5D-3L:** European Quality Of Life 5-Dimensions 3-Levels; **HDA:** high disease activity; **HLA-B27:** human leukocyte antigen B27; **HRQoL:** health-related quality of life; **hs:** high sensitivity; **ID:** inactive disease; **IL:** interleukin; **LDA:** low disease activity; **nr-axSpA:** non-radiographic axSpA; **PBO:** placebo; **Q4W:** every 4 weeks; **r-axSpA:** radiographic axSpA; **SD:** standard deviation; **SF-36 PCS:** Short-Form 36-Item Health Survey Physical Component Summary; **TNFi:** tumor necrosis factor inhibitor; **UK:** United Kingdom; **ULN:** upper limit of normal; **vHDA:** very high disease activity.

Institutions: [1]University Hospitals, Case Western Reserve University, Cleveland, OH, USA; [2]Division of Arthritis & Rheumatic Diseases, Oregon Health & Science University, Portland, OR, USA; [3]Swedish Medical Center and Providence St. Joseph Health, University of Washington, Seattle, WA, USA; [4]Department of Rheumatology, La Paz University Hospital, IdiPaz, Madrid, Spain; [5]Department of Rheumatology, Leiden University Medical Center, Leiden, The Netherlands; [6]University of Bielefeld, Klinikum Bielefeld, Bielefeld, Germany; [7]PCOM Analytics, Avallon, France; [8]UCB Pharma, Brussels, Belgium; [9]UCB Pharma, Colombes, France; [10]UCB Pharma, Copenhagen, Denmark; [11]UCB Pharma, Monheim am Rhein, Germany; [12]Division of Rheumatology, Department of Medicine, UMass Chan Medical School and UMass Memorial Medical Center, Worcester, MA, USA.
References: [1]Baraliakos X. Arthritis Rheumatol. 2022; 74 (suppl 9); [2]Boel A. Ann Rheum Dis 2019;78:1545–9. **Author Disclosures:** **MM**: Consultant for AbbVie, BMS, Eli Lilly, Novartis, Pfizer, and UCB Pharma; research grants from AbbVie, BMS, and UCB Pharma. **AD**: Speakers bureau from Janssen, Novartis and Pfizer; consultant for AbbVie, Amgen, Aurinia, BMS, Eli Lilly, Janssen, MoonLake, Novartis, Pfizer, and UCB Pharma; grant/research support from AbbVie, BMS, Celgene, Eli Lilly, MoonLake, Novartis, Pfizer, and UCB Pharma. **PJM**: Speakers bureau from AbbVie, Amgen, Eli Lilly, Janssen, Novartis, Pfizer, and UCB Pharma; consultant for AbbVie, Acerlyrin, Aclaris, Amgen, BMS, Boehringer Ingelheim, Eli Lilly, Galapagos, Gilead, GSK, Janssen, MoonLake, Novartis, Pfizer, Sun Pharma, and UCB Pharma; research grants from AbbVie, Amgen, BMS, Eli Lilly, Gilead, Janssen, Novartis, Pfizer, Sun Pharma, and UCB Pharma. **VNC**: Speakers bureau from AbbVie, Eli Lilly, Janssen, MSD, Novartis, Pfizer, and UCB Pharma; consultant for AbbVie, Eli Lilly, Galapagos, MoonLake, MSD, Novartis, Pfizer, and UCB Pharma; grant/research support from AbbVie and Novartis. **SR**: Consultant for AbbVie, Eli Lilly, Novartis, Pfizer, Sanofi, and UCB Pharma; research grants from AbbVie, Galapagos, MSD, Novartis, Pfizer, and UCB Pharma. **MR**: Speakers bureau from AbbVie, Boehringer Ingelheim, Chugai, Eli Lilly, Janssen, Novartis, Pfizer, and UCB Pharma; consultant for AbbVie, Eli Lilly, Novartis, and UCB Pharma. **CdlL**: Consultant for UCB Pharma. **CF, VT, MFM, UM**: Employees of UCB Pharma. **JK**: Consultant for Alvotech Swiss AG, Boehringer Ingelheim, Organon, Ridgeline Discovery, Samsung Bioepis, Sandoz Inc., Scipher Medicine, and UCB Pharma; grant/research support paid to institution from Gilead and Novartis. **Acknowledgments:** We would like to thank the patients and their caregivers in addition to all the investigators and their teams who contributed to this study. The authors acknowledge Celia Menckeberg, PhD, UCB Pharma, for publication coordination, Ellie Fung, BSc, Costello Medical, London, and Joseph Smith, PhD, Costello Medical, Cambridge, UK for medical writing and editorial assistance, and the Costello Medical Creative team for design support. This study was funded by UCB Pharma. All costs associated with development of this presentation were funded by UCB Pharma.

To receive a copy of this poster, scan the QR code or visit:
https://ucbposters.com/?PosterID=ISPOR2023_PCR68

Poster ID: ISPOR2023_PCR68
Link expiration: May 24th 2023