Treatment Patterns and Healthcare Resource Utilization in Patients With Relapsed/Refractory Multiple Myeloma (RRMM) on Second and Third Line (2L/3L) Therapy, Classified by Urbanicity and Ethnicity

Alexa Molinari, PharmD[1,2], Natalie Boytsov, PhD[1], Joseph Tkacz, MS[3], Peter Feng Wang, PhD[1], Sue Perera, PhD[4], Kris Norris[3], Suzanne Landi, PhD[1]*, Boris Gorsh, PharmD[1]

[1]GSK, Upper Providence, PA, USA; [2]Rutgers Center for Health Outcomes, Policy, and Economics, Rutgers, The State University of New Jersey, Piscataway, NJ, USA; [3]Inovalon, Bowie, MD, USA; [4]GSK, London, UK

*At the time of the study

Poster No. HSD57

Aims

Multiple myeloma (MM) is a hematologic malignancy characterized by uncontrolled proliferation of clonal plasma cells.[1]

Advances in treatment have improved outcomes for patients with relapsed/refractory MM (RRMM), with a 5-year survival of approximately 55%.[1–4] However, RRMM remains incurable and long-term clinical management is complex, with little standardization of treatment regimens.[4,5]

Disparities in mortality, incidence, and treatment utilization exist; some newer and more effective therapies for MM are underutilized, with barriers related to cost and access to treatment.[6]

In particular, ethnic minority groups can face barriers to treatment linked with access to care, and perception of and level of trust in healthcare professionals.[6]

To date, there is limited published real-world evidence for the RRMM patient population with respect to health disparities. A better understanding of disparities in healthcare resource utilization (HCRU) and treatment patterns is necessary to improve care-related decisions for these patients.

The Medicare fee-for-service (FFS) population includes most individuals in the US over 65 years of age, including approximately 60 million patients as of 2019,[7] and reflects the population of patients with RRMM in terms of key characteristics such as age and prevalence of comorbidities.[1] This population therefore represents an appropriate sample in which to explore real-world health expenditure and HCRU in RRMM.

Objectives

To describe treatment patterns and to measure direct healthcare expenditure and resource utilization in the second-line (2L) and third-line (3L) setting among patients with US Medicare coverage and a diagnosis of RRMM stratified by their urbanicity and ethnicity.

Methods

Study design

This was a retrospective, observational cohort analysis of adult patients with RRMM using administrative claims data from the US Medicare FFS claims database.

The analysis included patients who had previously received ≥1 prior line of MM therapy and who initiated 2L therapy between January 1, 2016, and December 31, 2020.

- The diagnostic index date, to confirm the diagnosis of MM, was the date of the earliest claim with an MM diagnosis code (minimum 6-month disease-naïve period required before diagnostic index).
- The treatment index date was the initiation of 2L treatment, marking the end of the baseline period and start of the follow-up period.

Patients were required to have a minimum of 6 months of continuous enrollment preceding the diagnostic index and no claims with MM diagnosis codes during this period.

Patients were also required to be continuously enrolled in the database for ≥12 months before and ≥30 days after the treatment index date.

Treatment patterns, HCRU, and expenditures were reported descriptively by line of therapy (2L or 3L), race (White, Black, or other), and location (urban or rural residence).

Results

Patient population

A total of 4560 patients initiated 2L treatment, of which 1991 (43.7%) subsequently initiated 3L treatment. The analysis by race and geographical residence included 4434 patients (97.2%) in the 2L cohort and 1941 patients (97.5%) in the 3L cohort, once patients with unknown race were excluded (**Table 1**).

Mean ± standard deviation (SD) age in overall the 2L cohort was 75.1 (7.9) years and 48.4% were male; similarly, in the overall 3L cohort mean age was 74.5 (7.7) years and 46.7% were male. Most patients were indexed between 2018 and 2020.

Table 1. Patient characteristics

	2L (N=4434)					3L (N=1941)				
	White (n=3651)	Black (n=622)	Other (n=161)	Urban (n=3178)	Rural (n=1256)	White (n=1613)	Black (n=267)	Other (n=61)	Urban (n=1370)	Rural (n=571)
Age, years, mean (SD)	75.8 (7.6)	71.9 (9.0)	74.8 (9.0)	75.3 (7.9)	74.9 (8.1)	75.2 (7.3)	70.9 (8.8)	74.2 (9.0)	74.7 (7.7)	74.1 (7.6)
Male, n (%)	1801 (49.3)	255 (41.0)	75 (46.6)	1517 (47.7)	614 (48.9)	764 (47.4)	109 (40.8)	27 (44.3)	629 (45.9)	271 (47.5)
Region,[a] n (%)										
Midwest	992 (27.2)	114 (18.3)	17 (10.6)	728 (22.9)	395 (31.4)	418 (25.9)	54 (20.2)	-	297 (21.7)	182 (31.9)
Northeast	683 (18.7)	108 (17.4)	28 (17.4)	695 (21.9)	124 (9.9)	312 (19.3)	50 (18.7)	-	310 (22.6)	62 (10.9)
South	1277 (35.0)	351 (56.4)	37 (23.0)	1116 (35.1)	549 (43.7)	575 (35.6)	142 (53.2)	12 (19.7)	494 (36.1)	235 (41.2)
West	696 (19.1)	45 (7.2)	79 (49.1)	637 (20.0)	183 (14.6)	306 (19.0)	18 (6.7)	32 (52.5)	268 (19.6)	88 (15.4)
Treatment index,[a] n (%)										
2016	561 (15.4)	108 (17.4)	26 (16.1)	478 (15.0)	217 (17.3)	330 (20.5)	62 (23.2)	17 (27.9)	280 (20.4)	129 (22.6)
2017	618 (16.9)	111 (17.8)	21 (13.0)	535 (16.8)	215 (17.1)	343 (21.3)	59 (22.1)	-	292 (21.3)	117 (20.5)
2018	734 (20.1)	117 (18.8)	31 (19.3)	619 (19.5)	263 (20.9)	371 (23.0)	62 (23.2)	17 (27.9)	319 (23.3)	131 (22.9)
2019	803 (22.0)	149 (24.0)	31 (19.3)	708 (22.3)	275 (21.9)	374 (23.2)	54 (20.2)	12 (19.7)	312 (22.8)	128 (22.4)
2020	935 (25.6)	137 (22.0)	52 (32.3)	838 (26.4)	286 (22.8)	195 (12.1)	30 (11.2)	-	167 (12.2)	66 (11.6)
Low-Income Subsidy, n (%)[b]	497 (13.6)	302 (48.6)	123 (76.4)	652 (20.5)	270 (21.5)	203 (12.6)	124 (46.4)	50 (82.0)	260 (19.0)	117 (20.5)
Follow-up, months, mean (SD)[c]	19.1 (14.8)	18.2 (14.3)	18.1 (15.5)	18.7 (14.7)	19.4 (14.8)	24.5 (14.6)	24.7 (14.3)	27.3 (15.2)	24.8 (14.6)	24.4 (14.5)

[a]Patient numbers <11 were masked per reporting requirements under the CMS data use agreement; [b]qualified for Low-Income Subsidy for Medicare prescription drug coverage; [c]from treatment index date
2L, second line; 3L, third line; CMS, US Centers for Medicare and Medicaid Services; SD, standard deviation

Of the 2L cohort, 82.3% were White and 14.0% Black, and 71.7% resided in an urban setting (**Figure 1**); in the 3L group, 83.1% were White and 13.8% Black, and 70.6% resided in urban areas.

Geographical residence varied by race, with a greater proportion of the Black and other race cohorts residing in urban settings compared with the White cohort.

Figure 1. Race and geographical residence among 2L patients (N=4434)

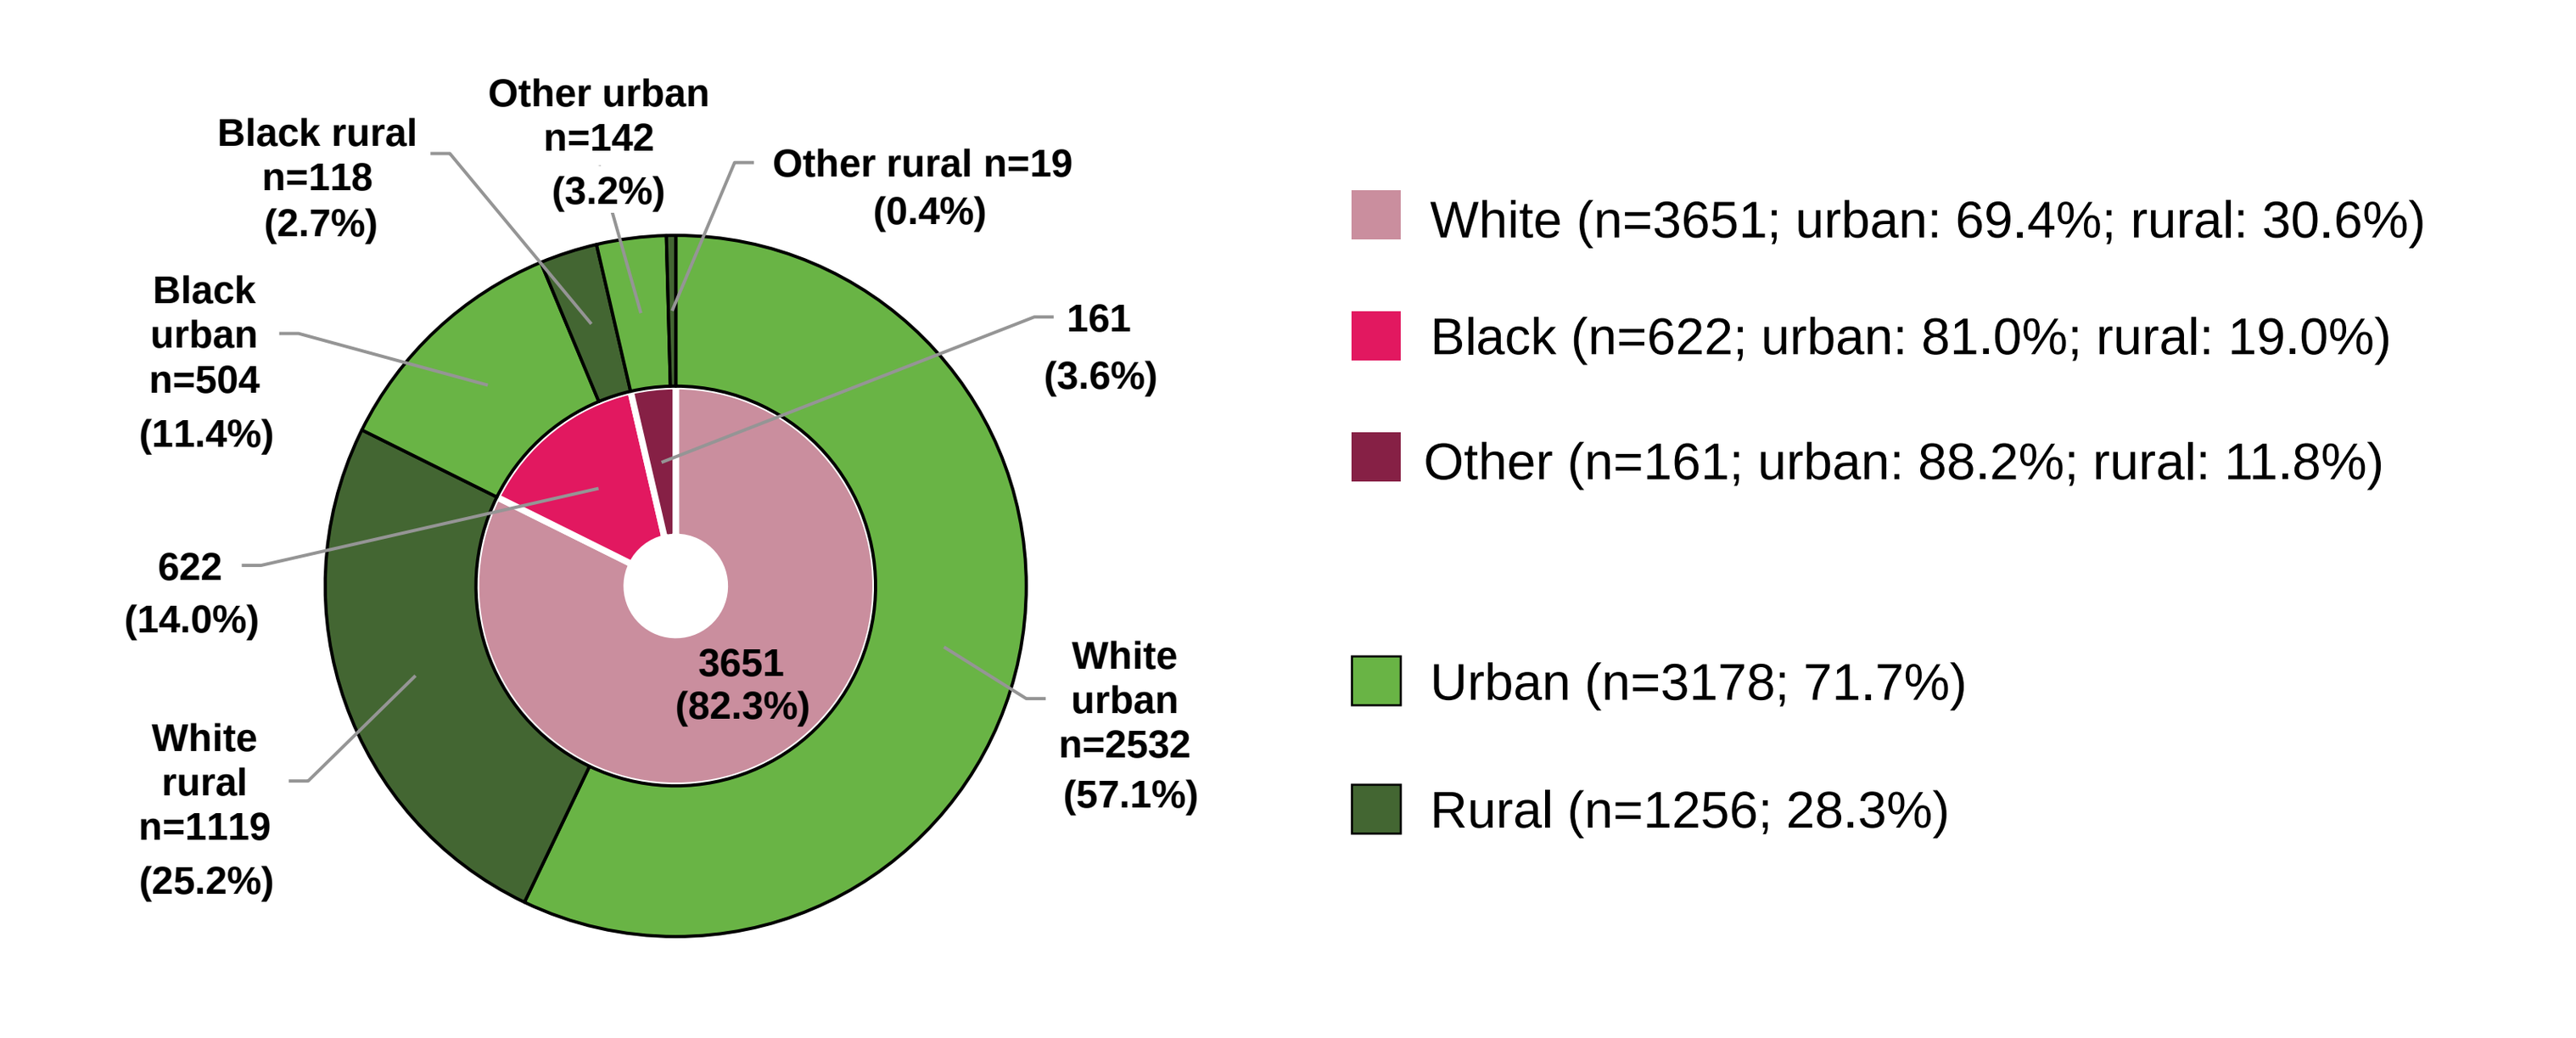

Black rural n=118 (2.7%)
Other urban n=142 (3.2%)
Other rural n=19 (0.4%)
Black urban n=504 (11.4%)
161 (3.6%)
622 (14.0%)
3651 (82.3%)
White urban n=2532 (57.1%)
White rural n=1119 (25.2%)
White (n=3651; urban: 69.4%; rural: 30.6%)
Black (n=622; urban: 81.0%; rural: 19.0%)
Other (n=161; urban: 88.2%; rural: 11.8%)
Urban (n=3178; 71.7%)
Rural (n=1256; 28.3%)

Other included native Hawaiian or other Pacific Islander, Hispanic or Latino, and American Indian and Alaska Native.
2L, second line

Treatment patterns

Treatment regimen utilization did not differ greatly, with the most frequently used agents (bortezomib, lenalidomide, and daratumumab) remaining similar between ethnicity subgroups (**Figure 2**).

Figure 2. The 5 most common treatment regimens used at 1L, 2L, and 3L treatment by race

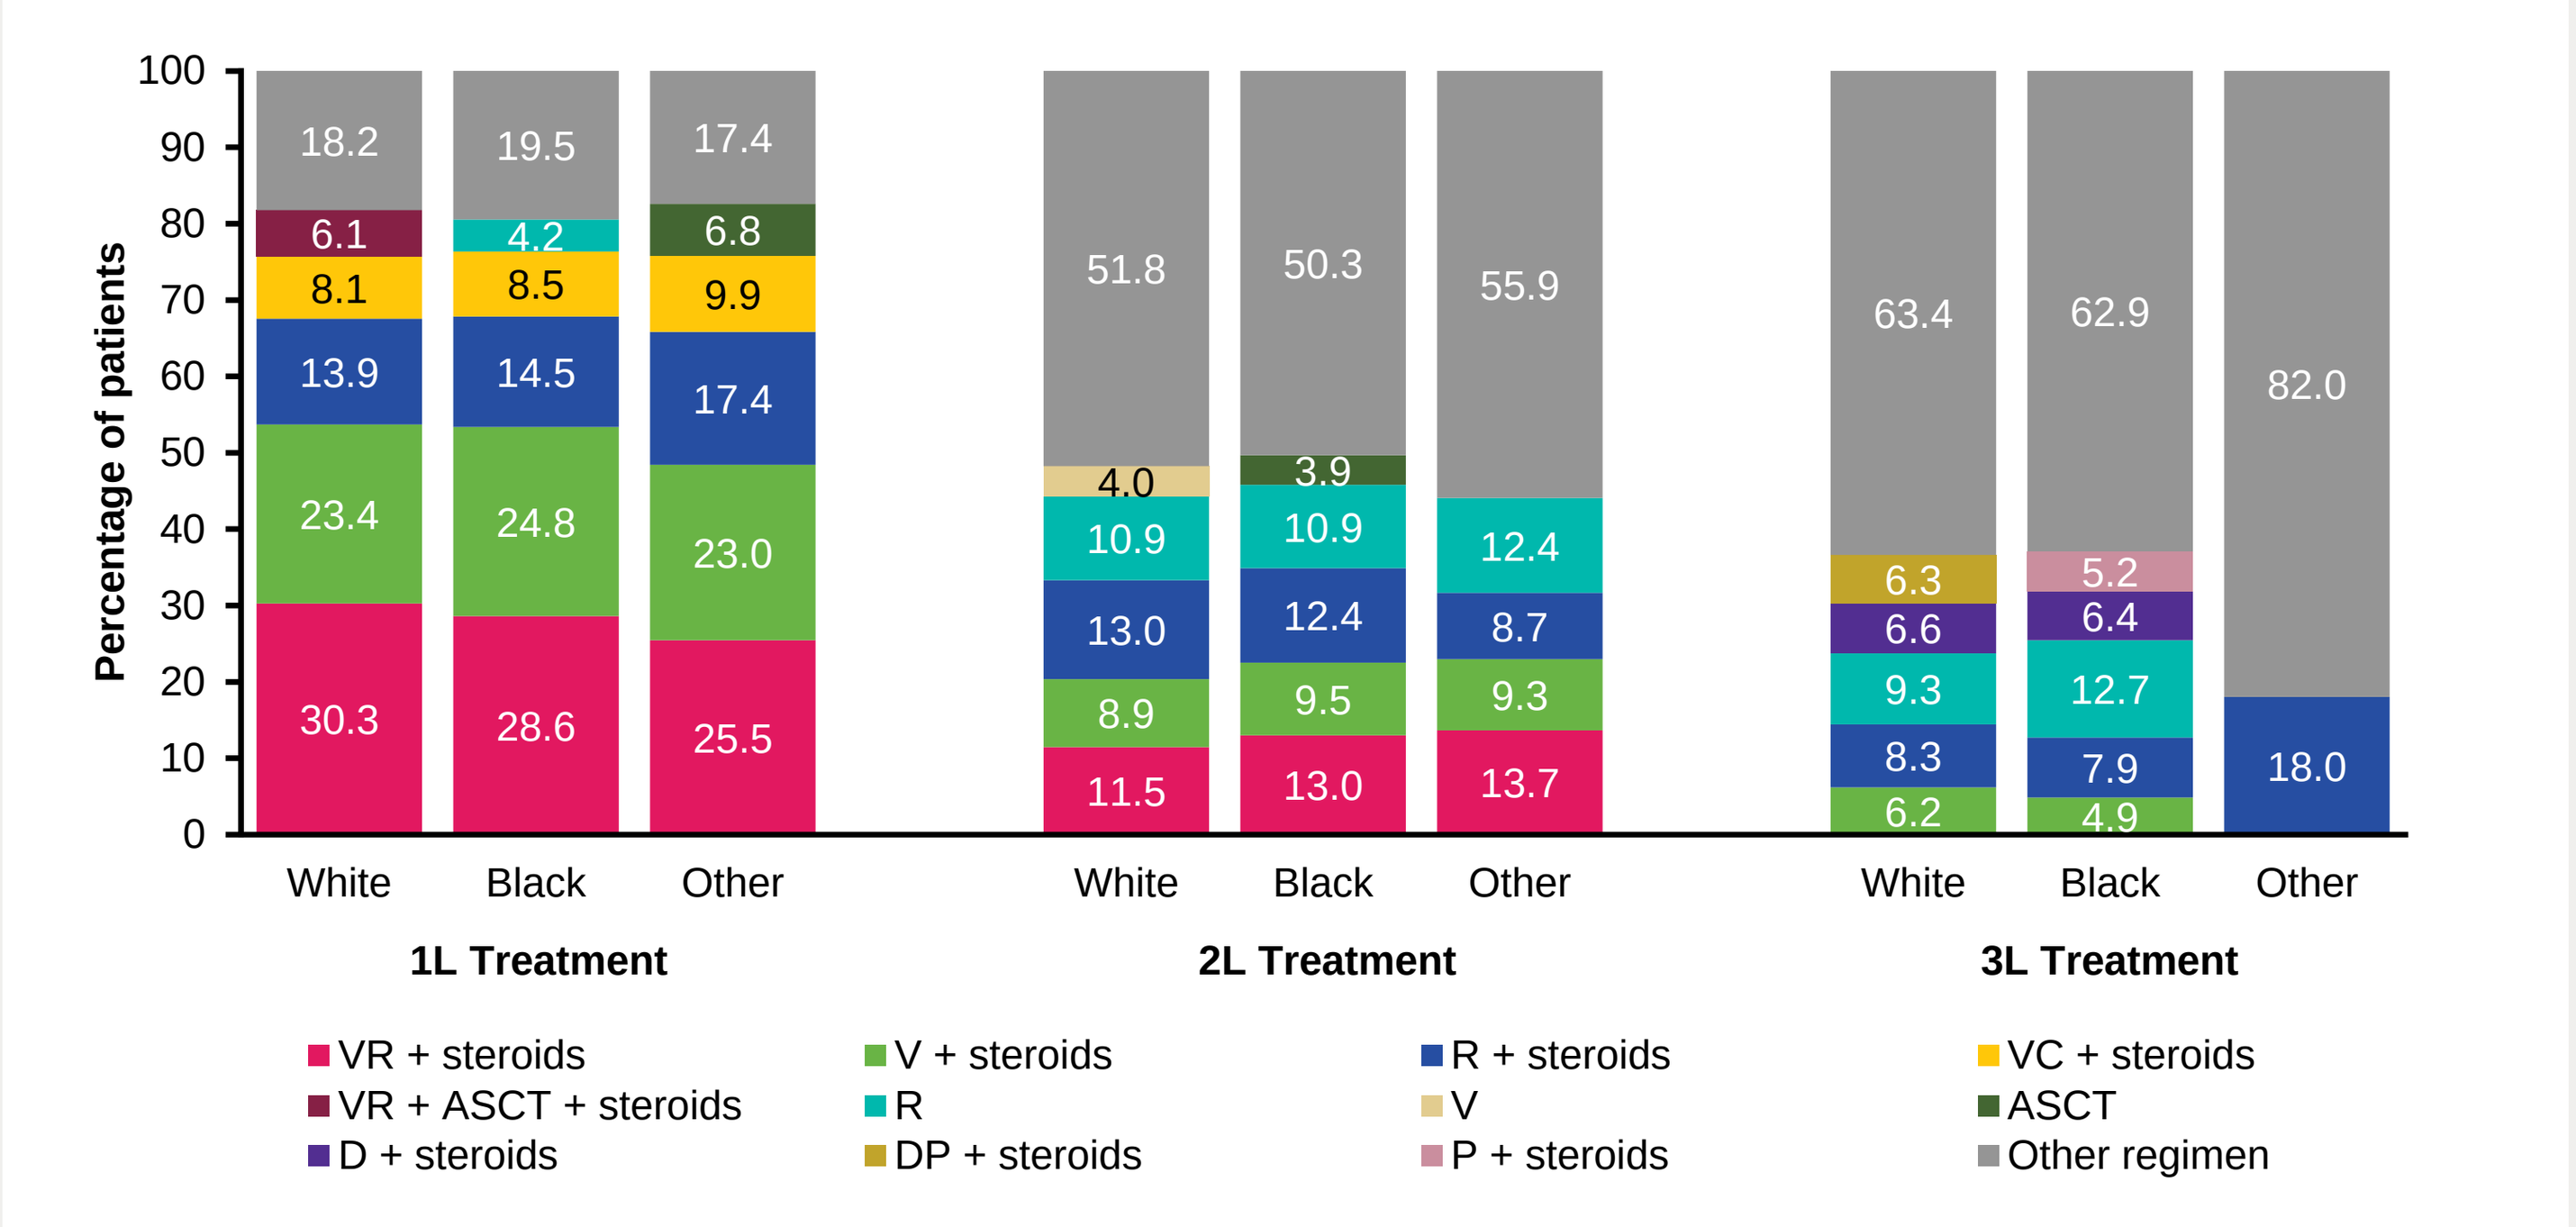

Percentage of patients
0 10 20 30 40 50 60 70 80 90 100
1L Treatment: White 30.3, 23.4, 13.9, 8.1, 6.1, 18.2; Black 28.6, 24.8, 14.5, 8.5, 4.2, 19.5; Other 25.5, 23.0, 17.4, 9.9, 6.8, 17.4
2L Treatment: White 11.5, 8.9, 13.0, 10.9, 4.0, 51.8; Black 13.0, 9.5, 12.4, 10.9, 3.9, 50.3; Other 13.7, 9.3, 8.7, 12.4, 55.9
3L Treatment: White 6.2, 8.3, 9.3, 6.6, 6.3, 63.4; Black 4.9, 7.9, 12.7, 6.4, 5.2, 62.9; Other 18.0, 82.0
VR + steroids
V + steroids
R + steroids
VC + steroids
VR + ASCT + steroids
R
V
ASCT
D + steroids
DP + steroids
P + steroids
Other regimen

The 5 most common treatment regimens are shown for each group; the remainder of patients received other regimens with lower frequency. Fewer than 5 different regimens were used by >11 patients (the threshold for masking under the CMS data use agreement) in 'Other' racial subgroups in 2L and 3L treatment.
1l, first line; 2L, second line; 3L, third line; ASCT, autologous stem cell transplant; C, cyclophosphamide; CMS, US Centers for Medicare and Medicaid Services; D, daratumumab; P, pomalidomide; R, lenalidomide; V, bortezomib

Figure 3. Frequency of ≥1 emergency department visit in the 2L (A) and 3L (B) cohorts

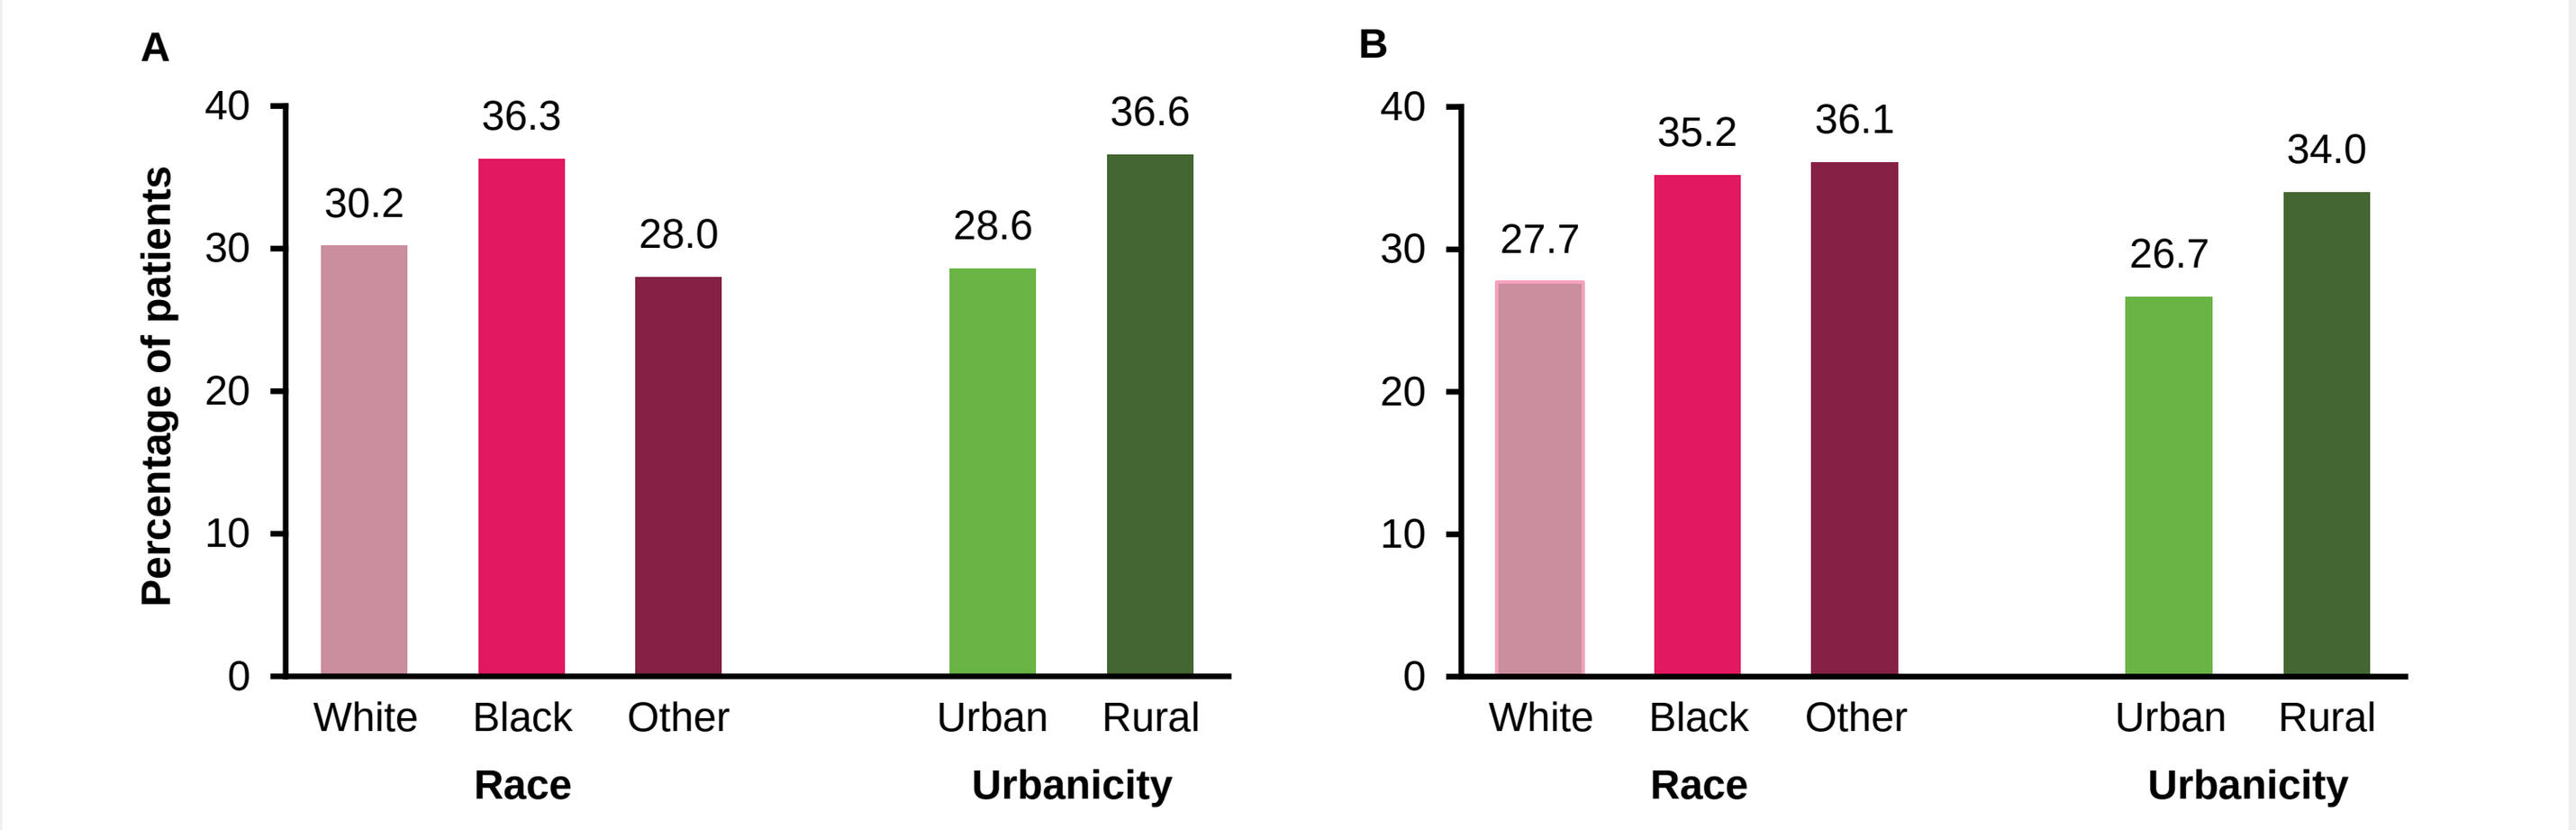

A
Percentage of patients
White 30.2
Black 36.3
Other 28.0
Urban 28.6
Rural 36.6
Race
Urbanicity
B
White 27.7
Black 35.2
Other 36.1
Urban 26.7
Rural 34.0
Race
Urbanicity

2L, second line; 3L, third line

Figure 4. Frequency of ≥1 inpatient hospital visit in the 2L (A) and 3L (B) cohorts

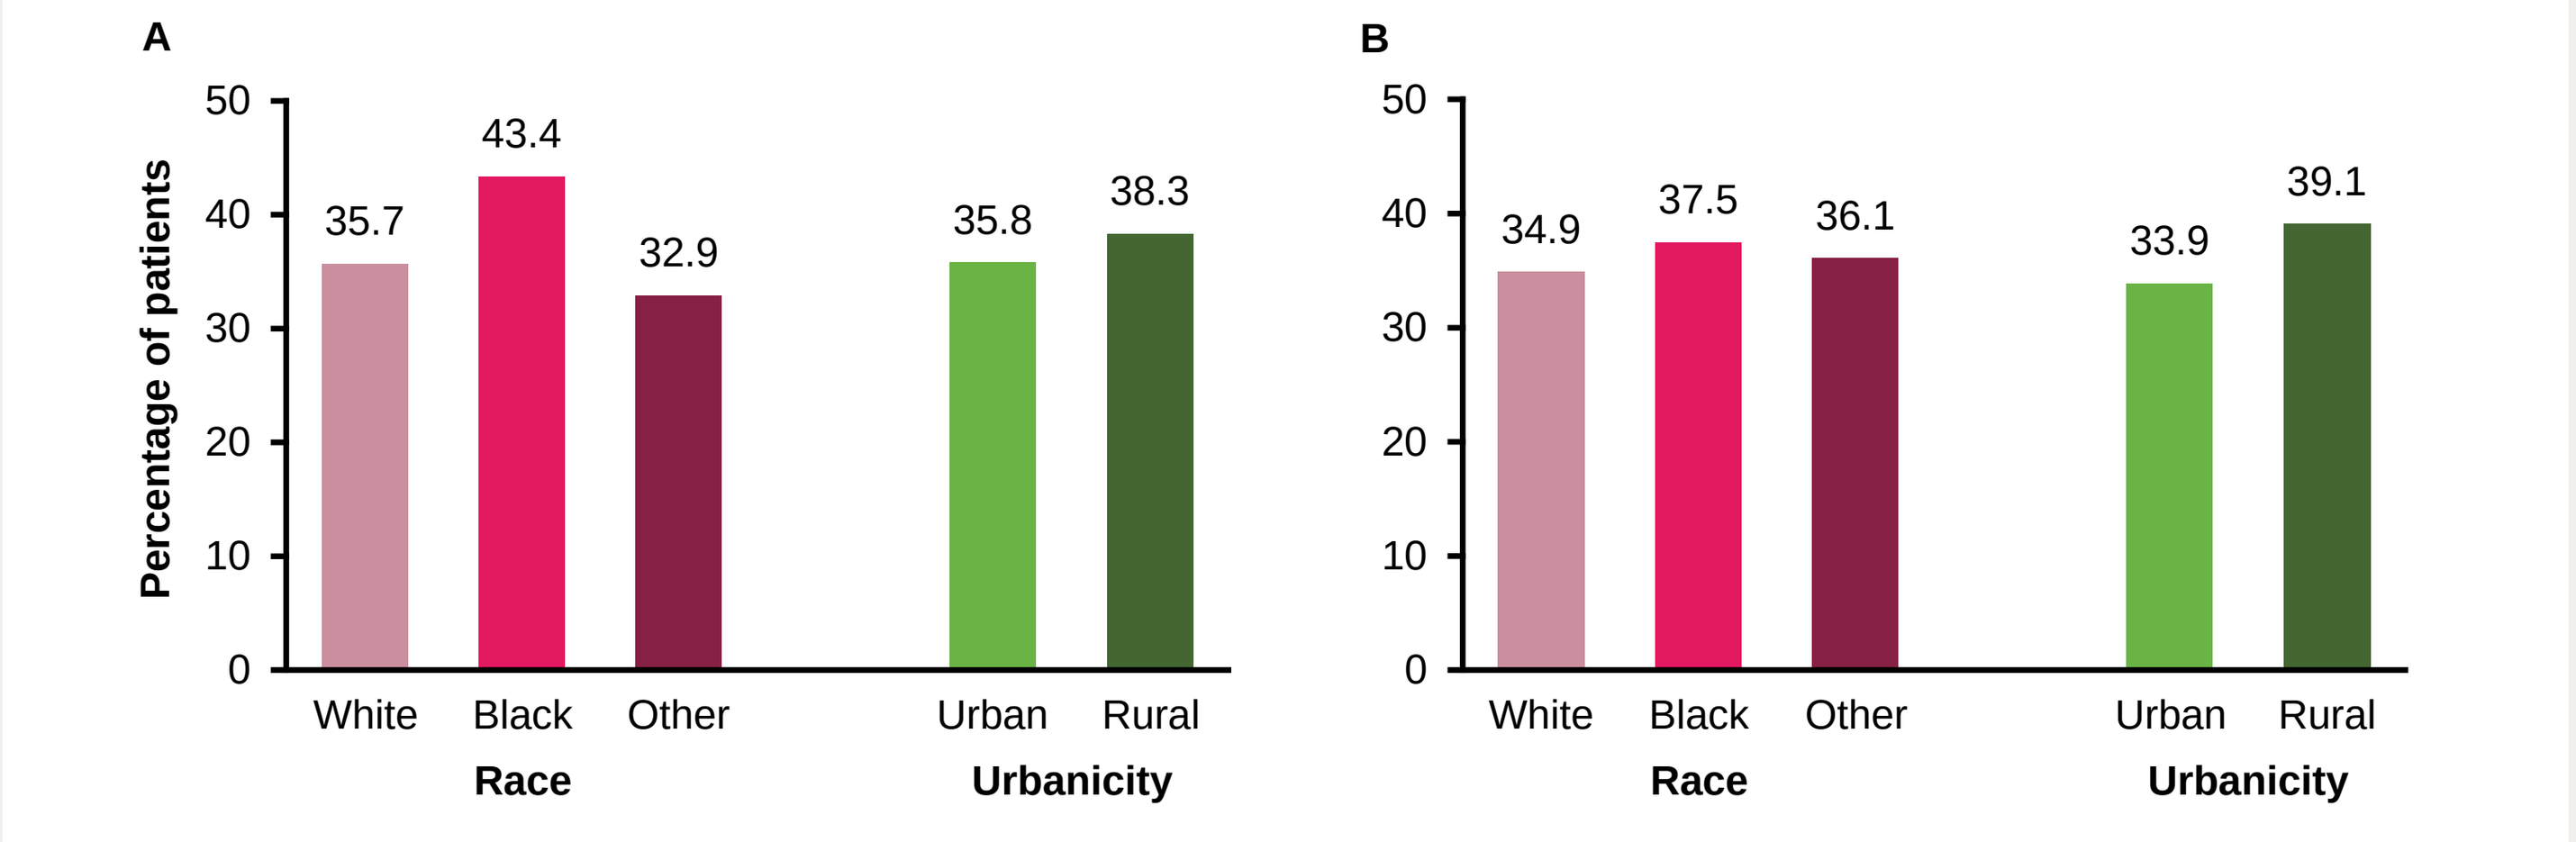

A
Percentage of patients
White 35.7
Black 43.4
Other 32.9
Urban 35.8
Rural 38.3
Race
Urbanicity
B
White 34.9
Black 37.5
Other 36.1
Urban 33.9
Rural 39.1
Race
Urbanicity

2L, second line; 3L, third line

HCRU

More Black patients presented for emergency care (≥1 emergency department [ED] visits) than White patients in 2L (36.3% vs 30.2%) and 3L (35.2% vs 27.7%) (**Figure 3**).

A larger proportion of Black patients also had received inpatient care (≥1 hospitalizations) than White patients in both 2L (43.4% vs 35.7%) and 3L (37.5% vs 34.9%) settings (**Figure 4**).

More patients in rural areas presented for emergency care and inpatient care than those in urban settings.

- In 2L, the proportion of patients with ≥1 ED visit was 36.6% in rural settings and 28.6% for those in urban areas; in 3L, the proportions were 34.0% and 26.7%, respectively.
- In patients initiating 2L treatment, the proportions of patients with ≥1 hospitalization were 38.3% in rural areas and 35.8% in urban areas; in 3L, the proportions were 39.1% and 33.9%.

Mean (SD) duration of hospitalizations in days remained consistent across race (White: 1.0 [3.1]; Black: 1.2 [3.0]; other: 1.2 [3.7]) and location subgroups (urban: 1.1 [3.3]; rural: 1.0 [2.5]).

Costs

Total (inpatient, outpatient, and pharmacy) costs per patient per month (PPPM) were similar across all groups.

- Pharmacy costs were the main contributor to total costs among all race subgroups in both 2L and 3L; pharmacy expenses were higher for 3L than 2L treatment in all race subgroups (**Figure 5**).
- Across all groups, inpatient costs exceeded outpatient costs for patients with ≥1 inpatient visit.

Cost differences between the urban and rural cohorts were similar in both 2L ($19,776 vs $19,226) and 3L settings ($21,455 vs $20,980).

The majority of HCRU and costs were attributable to MM-related care; pharmacy costs were the largest contributor to MM-related costs across all groups, followed by inpatient and outpatient costs.

Figure 5. Healthcare expenditure in 2L (A) and 3L (B) cohorts by race

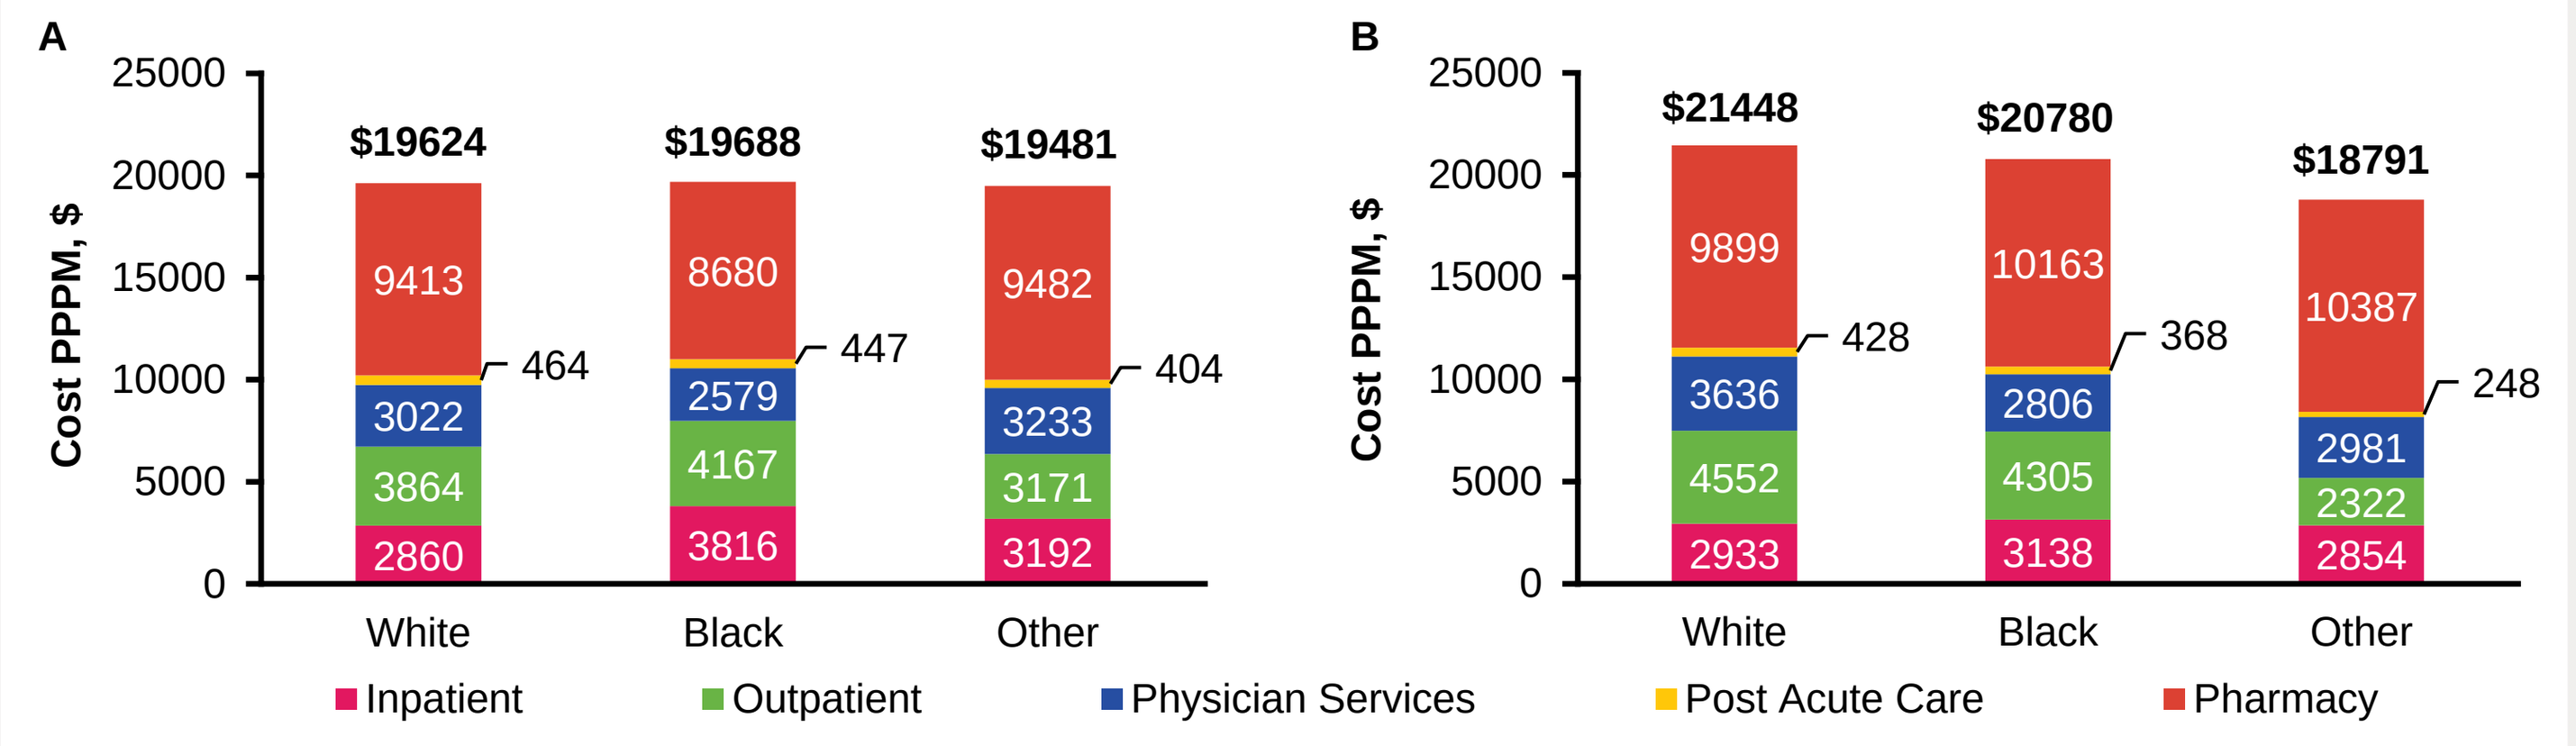

A
Cost PPPM, $
White $19624: 2860, 3864, 3022, 464, 9413
Black $19688: 3816, 4167, 2579, 447, 8680
Other $19481: 3192, 3171, 3233, 404, 9482
B
White $21448: 2933, 4552, 3636, 428, 9899
Black $20780: 3138, 4305, 2806, 368, 10163
Other $18791: 2854, 2322, 2981, 248, 10387
Inpatient
Outpatient
Physician Services
Post Acute Care
Pharmacy

2L, second line; 3L, third line; PPPM, per patient per month

Limitations

The study was limited to patients with Medicare FFS insurance and may not be generalizable to patients with other types of insurance or those who are uninsured.

There is potential for misclassification of variables such as medication usage and disease status. Medication usage was based on filled prescriptions and medical claims for physician-administered medications, and patients were assumed to have taken medications as prescribed.

Additionally, certain cohorts had small sample sizes, and the study design did not provide insight on the reasons for differences in HCRU between cohorts.

Conclusions

Limited variation in treatment patterns was observed by line of therapy, race, and urban or rural setting.

Emergency and inpatient care services were used by a higher proportion of Black patients than White patients, and by patients in rural compared with urban settings.

Despite disparities in HCRU, total costs were similar between groups; most costs were MM-related, with pharmacy-related costs accounting for the greatest proportion, followed by outpatient charges.

Despite the limited scope of this analysis, the findings highlight a need for further research to inform and improve care-related decisions and inform the community of additional actions needed to minimize differences in care and access.

Disclosures

AM is an employee of Rutgers, The State University of New Jersey, and is contracted with GSK. **NB**, **PFW**, **SP**, **SL**, and **BG** are employees of GSK and own stocks and shares in GSK. **JT** and **KN** are employees of Inovalon, which received research funding from GSK.

Acknowledgments

This study was funded by GSK (209838).

Editorial assistance was provided by Mark Condon, DPhil, of Fishawack Indicia Ltd, part of Fishawack Health, and funded by GSK. Programming support was provided by William Johnson (formerly of Inovalon).

References

1. Gatopoulou X, et al. *Clinicoecon Outcomes Res* 2022;14:731–53.
2. Cancer.Net. https://www.cancer.net/cancer-types/multiple-myeloma/statistics. Accessed March 2023.
3. Sonneveld P. *Hematology Am Soc Hematol Educ Program* 2017;508–17.
4. Bhatt P, et al. *Curr Oncol* 2023;30(2):2322–47.
5. Hernández-Rivas J, et al. *Biomark Res* 2022;10(1):1.
6. Fiala MA, et al. *Cancer* 2017;123(9):1590–6.
7. Kaiser Family Foundation. https://www.kff.org/medicare/issue-brief/an-overview-of-medicare/. Accessed March 2023.

Presented at ISPOR, Boston, MA, USA, May 7–10, 2023