EE7

The Importance of Considering Price Metrics and Market Dynamics in US Economic Evaluations: The Case of SGLT-2 Inhibitors in the Treatment of Type 2 Diabetes (T2D)

Michael Willis,[1] Cheryl Neslusan,[2,*] Andreas Nilsson[1]

[1]The Swedish Institute for Health Economics, Lund, Sweden; [2]Janssen Scientific Affairs, LLC, Titusville, NJ, USA.

*Presenting author.

An electronic version of the poster can be viewed by scanning the QR code.

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

OBJECTIVE

Illustrate the sensitivity of value estimates to price dynamics and price metrics using the case of the sodium glucose co-transporter 2 (SGLT-2) inhibitor canagliflozin 300 mg versus the dipeptidyl peptidase-4 (DPP-4) inhibitor sitagliptin 100 mg in 3rd-line treatment of type 2 diabetes mellitus

BACKGROUND

- Drug prices are dynamic (especially in the US setting)
 - Even while patent protected, manufacturers face within- and across-class competitive pressure from other compounds (varying over time, with new competitors and existing competitors losing exclusivity)
 - Generic entry into the market
 - IMS Health found average US drug price declines of 51% after 1 year and 77% after 10 years of becoming generic from 2002 to 2014[1]
 - New evidence can improve understanding of benefit-risk ratios of alternative treatments
 - Changes in marketplace structure and rules can alter buyer and seller bargaining power (e.g., pharmacy benefit manager [PBM] consolidation, the Inflation Reduction Act [IRA])
- Drug prices can be measured with multiple metrics
 - Economic evaluations routinely use *list prices* even though they ignore common price concessions (e.g., manufacturer rebates), and these prices rarely reflect actual transaction prices[2]
 - *Net prices* deduct these rebates and measure what manufacturers actually receive. However, they do not reflect true "economic costs" for society as they exclude the share of rebates that are retained by intermediaries (e.g., PBMs, pharmacies) for supply chain services[2]
 - *Systemwide net expenditure* (SNE), a term coined by Van Nuys et al, measures the "net price to manufacturers plus gross profits of all distribution system participants" (including intermediaries), reflecting the societal transaction price.[2] Calculating SNE requires confidential information and thus must be estimated
- The International Society for Pharmacoeconomics and Outcomes Research (ISPOR) Task Force on Good Research Practices for Measuring Drug Costs in Cost Effectiveness Analyses[3] recommends incorporating price dynamics and using appropriate price metrics, specifically prices that:
 - Account for time trends
 - Account for the presence (or anticipated presence) of generic alternatives
 - Are actually paid by a relevant stakeholder (e.g., SNE from the societal perspective, SNE less patient-borne costs from the insurer perspective, and patient-borne costs from the patient perspective)
- Most empirical research has neglected these recommendations, instead fixing drug prices at baseline and using list prices
 - In 2023, Schöttler et al[4] claimed to be the first to consider branded competition, with future drug price trajectories estimated via nonlinear regression equations. This approach requires access to retrospective price data, which would not exist for novel therapies
 - In a sample of US cost-effectiveness studies published before 2019 from the Tufts Medical Center Cost-Effectiveness Analysis (CEA) Registry, Neumann et al[5] found that only 5% even considered the impact of generic entry, the timing of which is reasonably predictable. Empirical work exists to support simulating price trajectories following generic entry.[6,7] A recent US study[8] illustrated several ways future generic prices might be applied in practice
 - Neumann et al[5] also found that 92% of studies in their sample used list prices and only 8% used net prices. To our knowledge, no studies have used SNE

METHODS

- Health outcomes and costs associated with these treatment strategies were simulated starting in 2014 (i.e., rolling back the clock to the launch of canagliflozin), enabling consideration of actual prices that evolved, alternative methods to predict these actual price trajectories that would have been available at the time, and alternative price metrics
- Economic simulation model
 - We used the Economic and Health Outcomes Model of Type 2 Diabetes Mellitus (ECHO-T2DM), a stochastic microsimulation model developed using Markov health states that captures microvascular and macrovascular complications
 - ECHO-T2DM allows for simulation of anti-hyperglycemic treatment strategies, associated biomarker changes, and adverse events and can account for the direct cardioprotective effects associated with SGLT-2 inhibitors.[9] It was adapted to support inclusion of year-specific prices. A full description is available[10-12]
- Key modeling assumptions
 - Baseline patient characteristics are presented in **Table 1**. Both groups were assumed to start therapy after failing to maintain HbA1c <7.0% while being treated with metformin and sulfonylurea
 - First-year biomarker changes and side-effect rates were sourced from a head-to-head randomized controlled trial,[13,14] and direct cardioprotective effects were sourced from the CANagliflozin cardioVascular Assessment Study (CANVAS) Program.[10] Conservatively, evidence from the Canagliflozin and Renal Events in Diabetes with Established Nephropathy Clinical Evaluation (CREDENCE) trial[15] was not incorporated in these simulations
 - Insulin was initiated as needed to maintain HbA1c <7.0%[16]
 - Costs for health complications were sourced from the literature[17,18]

Table 1. Baseline Patient Characteristics Used in Modeling

	Value*
Demographics	
Age, y	58.35 (11.37)
Males, %	55.1
Disease duration, y	9.6 (6.16)
Clinical indicators	
Smoker	0.05
HbA1c, %	7.84 (1.60)
SBP, mmHg	130.70 (13.57)
BMI, kg/m^2	31.60 (6.91)
eGFR, mL/min/1.73 m^2	91.62 (21.33)
Presence of comorbidities at baseline, %	
Macrovascular	
IHD (not including MI)	18.0
MI	3.0
CHF	5.0
Stroke	2.1

SBP, systolic blood pressure; BMI, body mass index; eGFR, estimated glomerular filtration rate; IHD, ischemic heart disease; MI, myocardial infarction; CHF, congestive heart failure.
*Data are mean (standard deviation) unless otherwise noted. Estimates were obtained from Optum® data where available (results on file) and supplemented with trial data.

- Two sets of analyses addressing different questions were performed
 - Analysis 1
 - The aim was to compare 8-year drug costs estimated with the conventional fixed list price assumption with estimates based on actual observed list prices and with estimates based on alternative price metrics, leveraging actual price data for the period from 2014 to 2021
 - ◊ To illustrate the magnitude of error associated with the fixed price assumption, results using list prices fixed at their 2014 values were compared to those using actual time-varying list prices
 - ◊ To illustrate the implications of ignoring the time-varying nature of prices and of not using a price metric that best matches the perspective of the decision problem, results using list prices fixed at their 2014 values were compared to those using (1) actual net prices, (2) fixed net prices, (3) actual SNEs, and (4) fixed SNEs
 - Analysis 2
 - The aim was to illustrate the impact of price dynamics, including hypothetical generic entry and alternative price metrics, on some key economic outcomes for a chronic disease like type 2 diabetes mellitus (25-year incremental drug costs and incremental total costs) from the perspective of a researcher at the time of launch
 - ◊ To illustrate the impact of ignoring future price dynamics, results using list prices fixed at their 2014 values were compared to 2 alternative projected list price trajectories
 - ◊ To illustrate the impact of considering future price dynamics and alternative price metrics, results using list prices fixed at their 2014 values were compared to 2 alternative projected net price trajectories and 2 alternative projected SNE trajectories
 - ◊ To illustrate the impact of choosing alternative price metrics, results using list prices fixed at their 2014 values were compared to those using net prices fixed at their 2014 values and SNEs fixed at their 2014 values
- Drug prices
 - Three sets of list price, net price, and SNE trajectories were used to inform these analyses (9 trajectories in total). They are depicted in **Figure 1**
 1. Actual list and net prices and estimated SNEs (2014-2021)
 - ◊ List and net prices (which account for discounts including commercial and Medicaid rebates and manufacturer coupons) were obtained from the SSR Health database
 - ◊ SNEs were derived by applying the relationship between list prices, net prices, and SNE trajectories for insulin as reported by Van Nuys et al[2] to list and net prices for canagliflozin 300 mg and sitagliptin 100 mg
 2. Generic-entrance–adjusted list and net prices and SNE projections (2014-2039)
 - ◊ Fixed 2014 list and net prices and SNEs (see #1 above) held constant until 2027 for sitagliptin and 2031 for canagliflozin, when generic entrance to the market is assumed to occur. These dates were sourced from Drugpatentwatch.com
 - ◊ Drug prices were assumed to decline to 63% of the price prior to generic entry during the 1st year and to 50% during the 2nd and subsequent years[6]
 3. Generic-entrance– and time-trend–adjusted list and net prices and SNEs (2014-2039)
 - ◊ Same as #2 in terms of generic entry, but with additional price dynamics considered. Logarithmic forecast equations based on actual DPP-4 prices and calculated SNEs from 2007 to 2014 were used to project list and net prices and SNEs over the period 2015 to 2039

Figure 1. Price trajectories for canagliflozin and sitagliptin by price metric and time-trend assumptions.

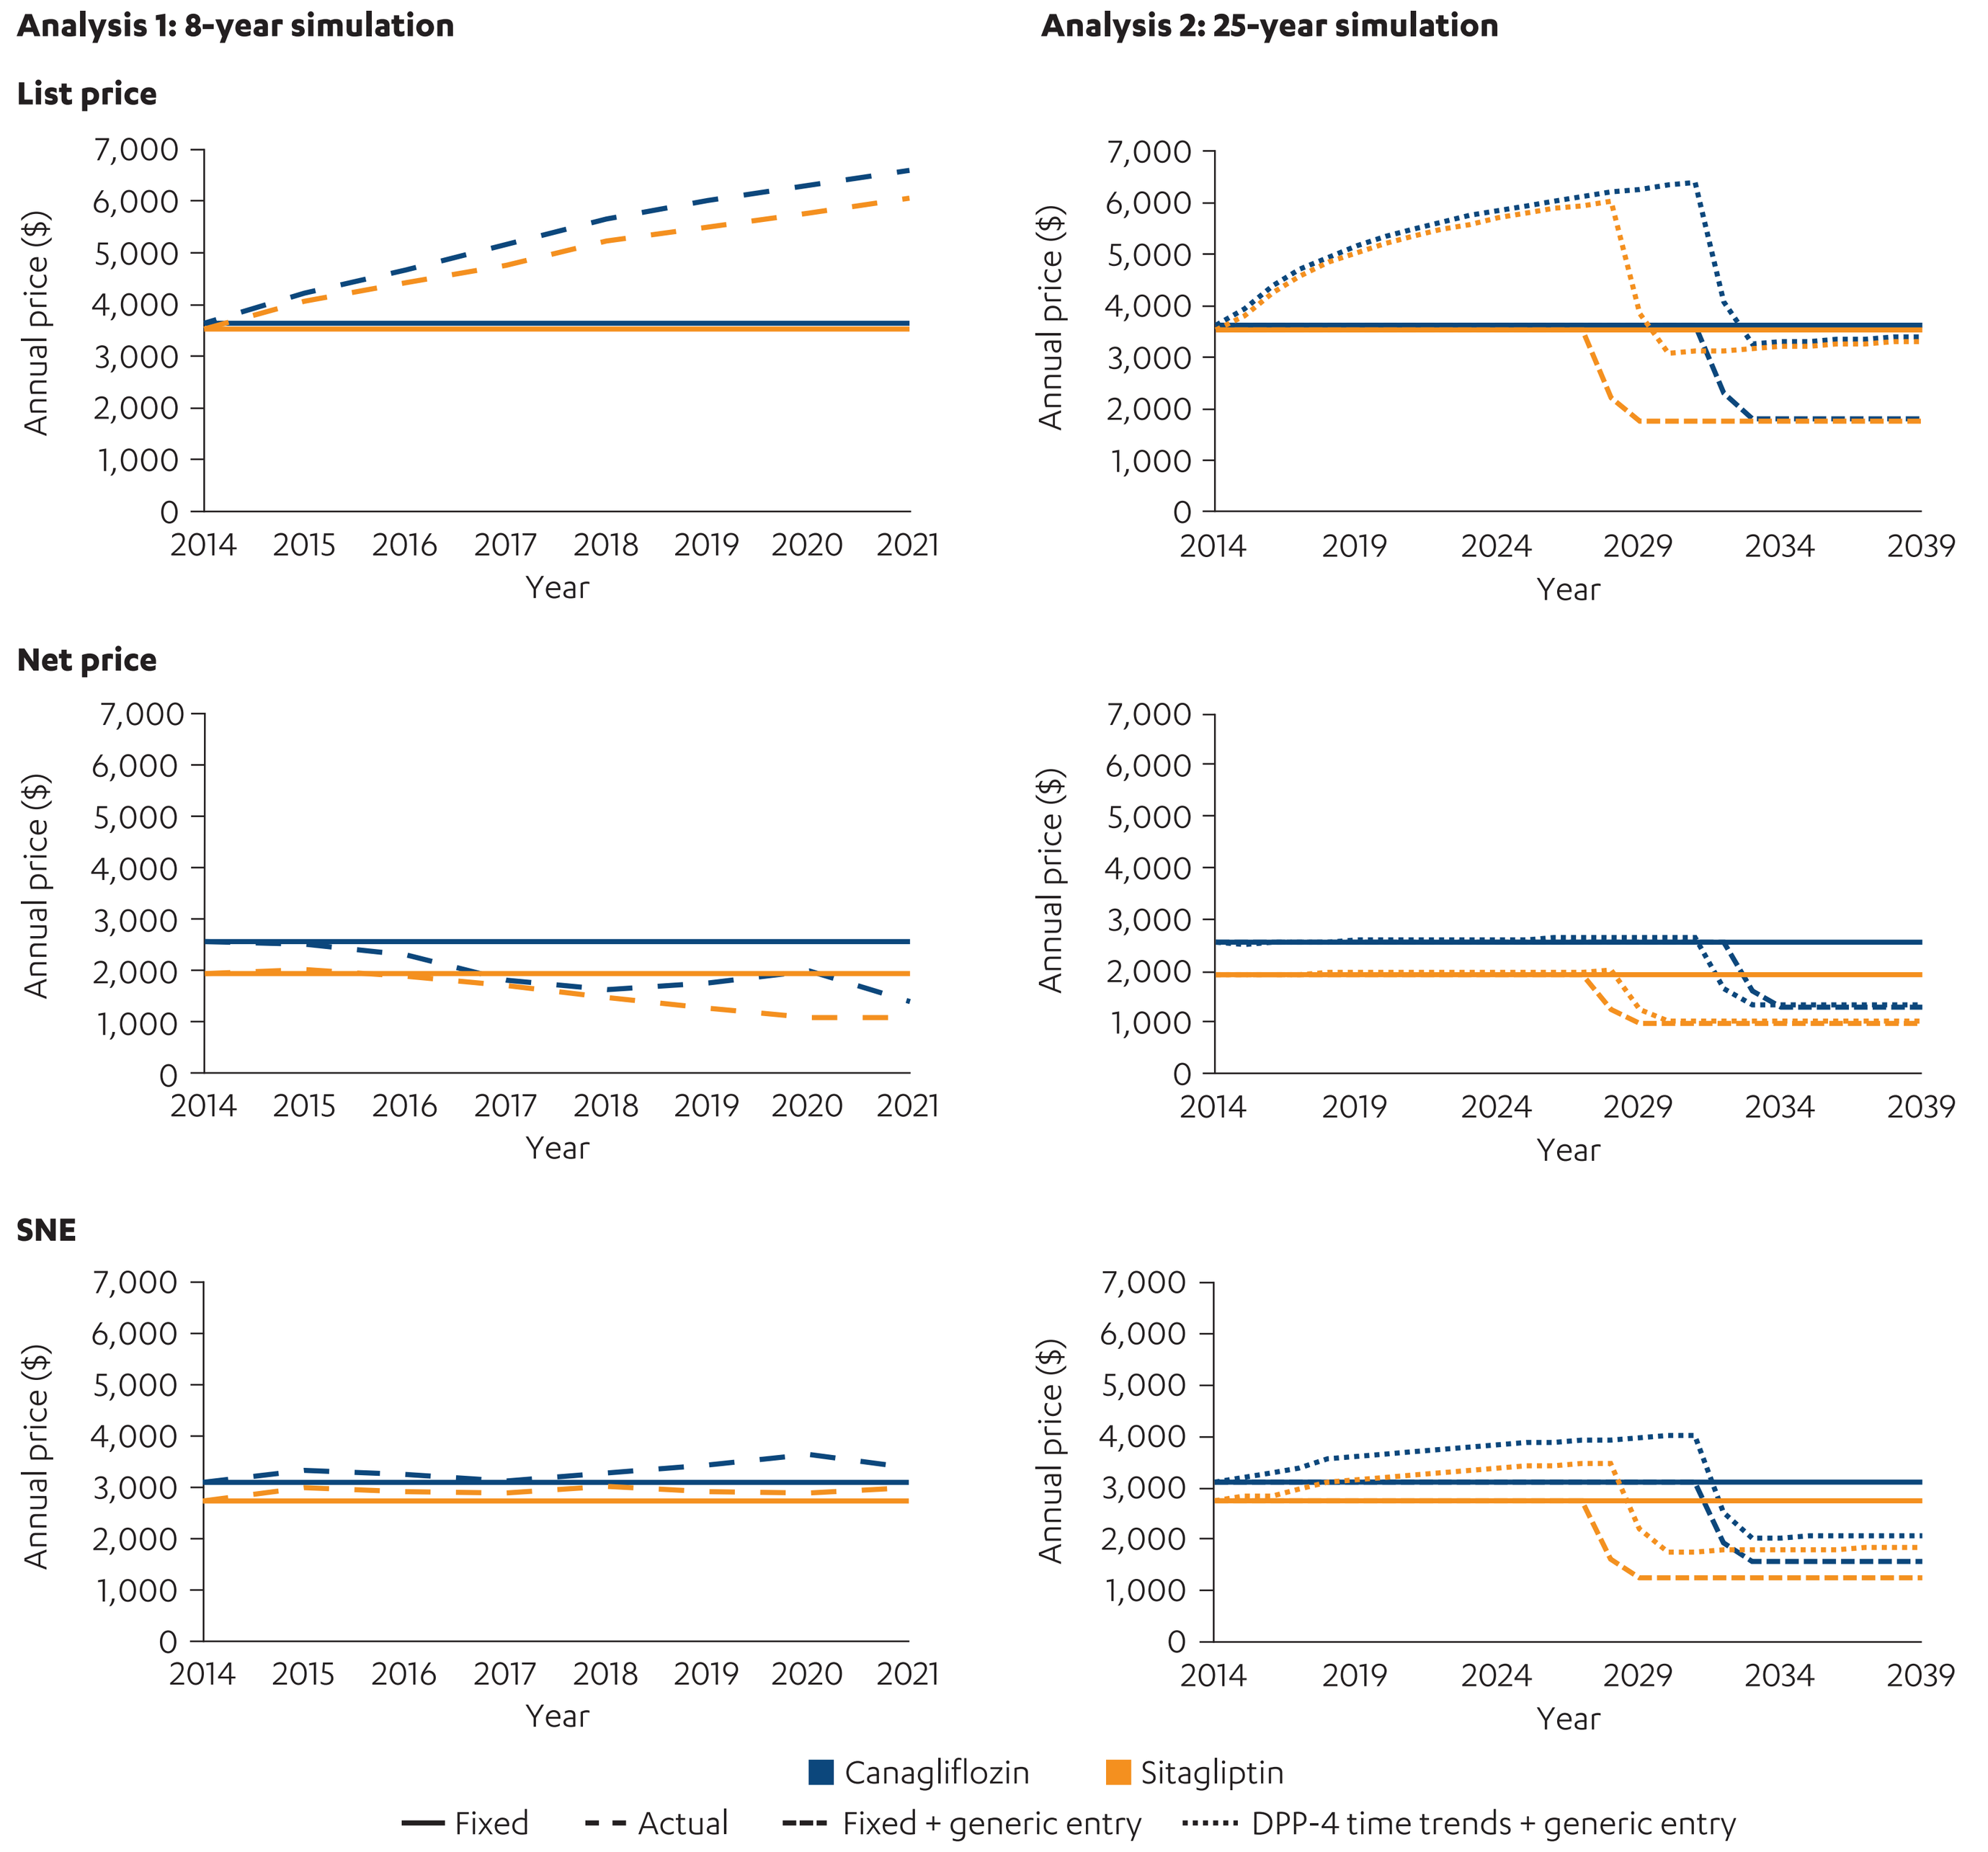
Analysis 1: 8-year simulation
Analysis 2: 25-year simulation
List price
Net price
SNE
Annual price ($)
Year
Canagliflozin
Sitagliptin
Fixed
Actual
Fixed + generic entry
DPP-4 time trends + generic entry

SNE, systemwide net expenditure; DPP-4, dipeptidyl peptidase-4.

RESULTS

- As in previous studies,[14,19,20] canagliflozin was associated with fewer microvascular and macrovascular complications, resulting in sizable cost offsets and greater longevity (results available upon request)
- Analysis 1: Impact of the conventional fixed list price assumption on estimated drug costs (see **Figure 2**)
 - Using fixed list prices, estimated 8-year drug costs for canagliflozin and sitagliptin were similar ($22,108 and $22,102, respectively)
 - Estimated drug costs based on fixed list prices were 40% lower for canagliflozin and 35% lower for sitagliptin than estimates based on the actual list prices
 - Estimated drug costs based on fixed list prices were substantially greater than estimates based on net prices for both canagliflozin and sitagliptin (differing by 30% and 45%, respectively, when using fixed net prices and by 45% and 55% when using actual net prices). The effects were larger for sitagliptin, which increased incremental drug costs
 - Estimated drug costs based on fixed list prices were about 10% to 20% larger than estimates based on fixed and actual SNE for both drugs
- Analysis 2: Impact of price dynamics, including hypothetical generic entry and alternative price metrics, on incremental drug and incremental total costs from the perspective of a researcher at time of launch (see **Figure 3**)
 - Using fixed list prices, estimated 25-year incremental drug costs and incremental total costs associated with canagliflozin (vs sitagliptin) were –$1,711 and –$9,032, respectively (i.e., both types of costs were lower for the canagliflozin arm)
 - Estimated incremental drug costs and incremental total costs were $1,619 and –$5,702, respectively, when fixed list prices were adjusted for generic entry (assumed earlier for sitagliptin)
 - Estimated incremental drug costs and incremental total costs were $920 and –$6,400, respectively, when fixed list prices accounted for both time trends and hypothetical generic entry
 - Using net prices, which account for manufacturer rebates but exclude retained intermediary earnings, resulted in estimated incremental drug costs >0. Incremental total costs reflect cost offsets
 - SNEs, which best match societal transaction cost, resulted in estimated drug cost offsets and total cost savings for canagliflozin for each of the 3 price trajectories considered (fixed SNE, only generic-entrance–adjusted, and generic-entrance– and time-trend–adjusted)
 - The primary driver of the difference between incremental drug and total costs in all scenarios is the reduced need for insulin rescue therapy, with additional cost offsets provided by reductions in microvascular and macrovascular complications that are avoided with canagliflozin

Figure 2. Total drug costs for canagliflozin 300 mg and sitagliptin 100 mg over an 8-year period by price metric and time-trend assumption.

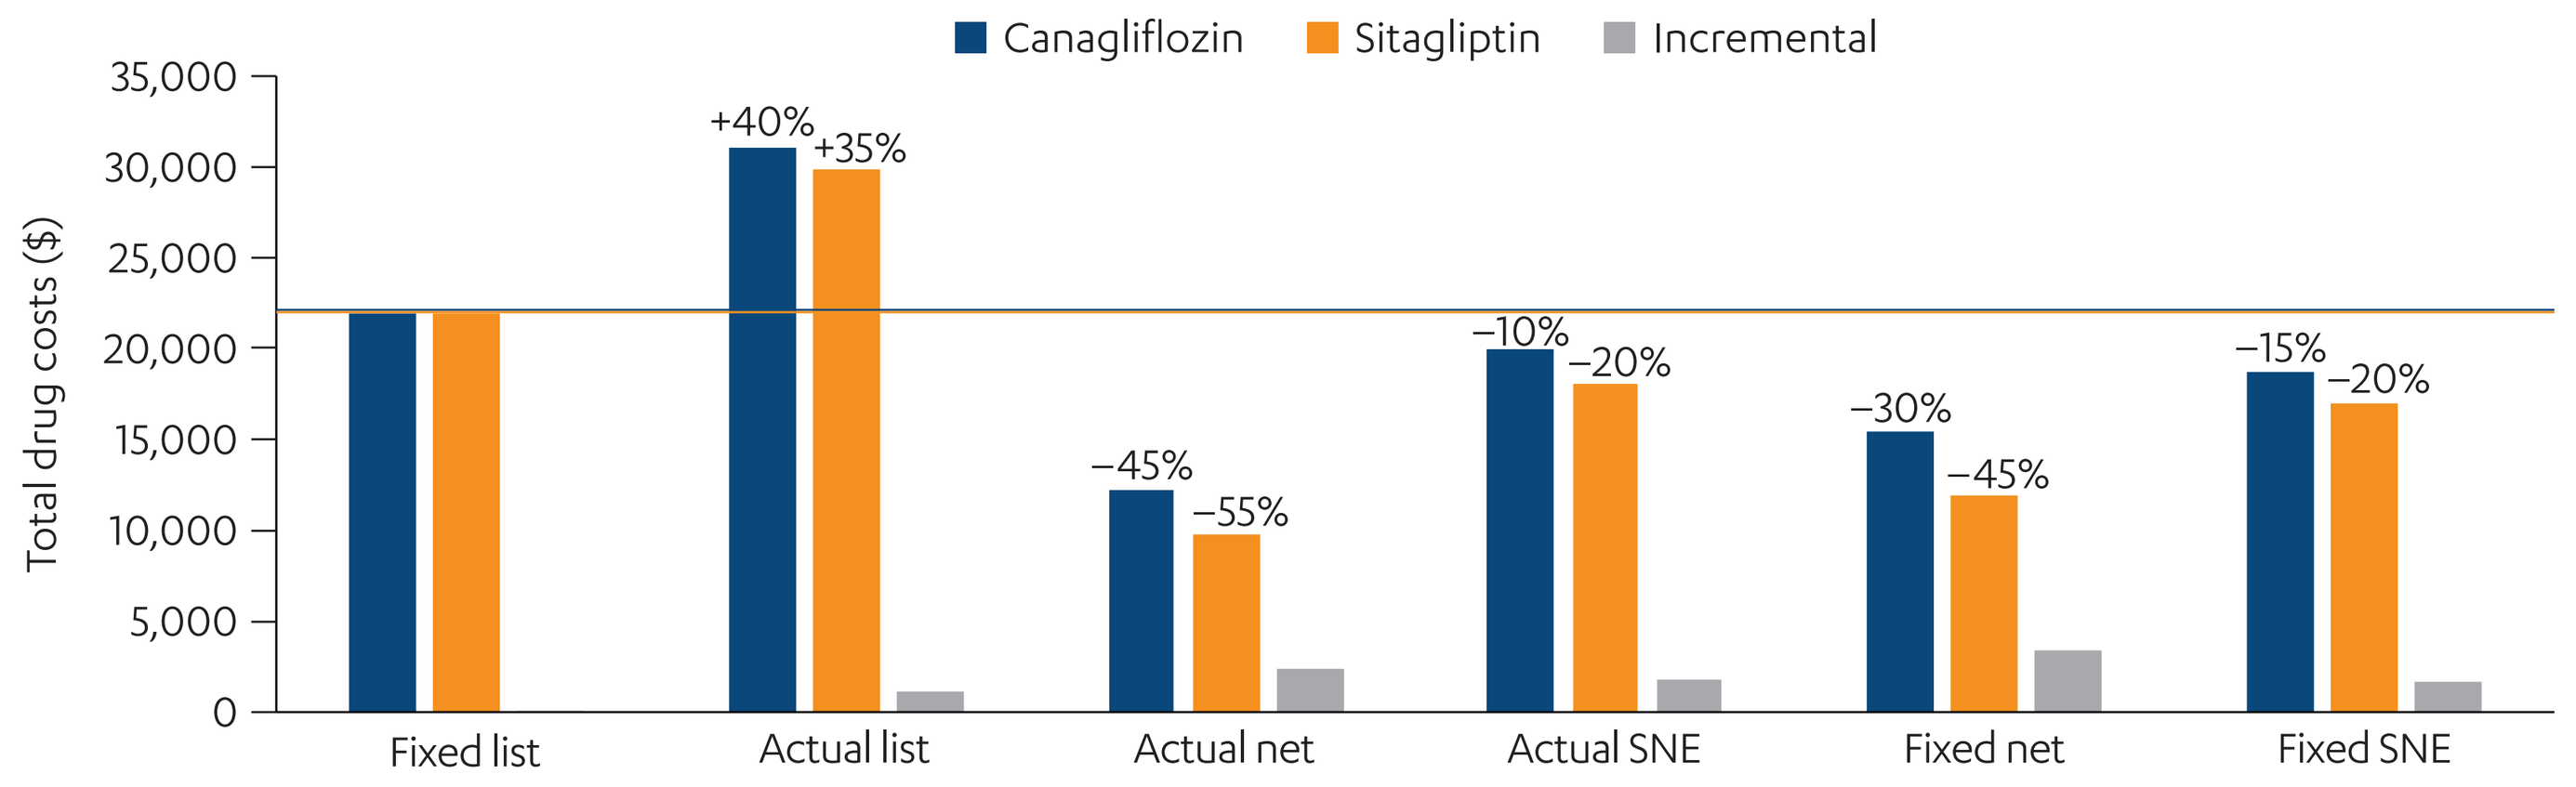
Canagliflozin
Sitagliptin
Incremental
Total drug costs ($)
+40%
+35%
−45%
−55%
−10%
−20%
−30%
−45%
−15%
−20%
Fixed list
Actual list
Actual net
Actual SNE
Fixed net
Fixed SNE

SNE, systemwide net expenditure.
Lines represent the fixed list price scenario. Reported percent differences are relative to conventional assumption of fixed 2014 list prices.

Figure 3. Incremental drug and incremental total costs for canagliflozin 300 mg versus sitagliptin 100 mg over 25 years by price metric and future price trajectory assumption.

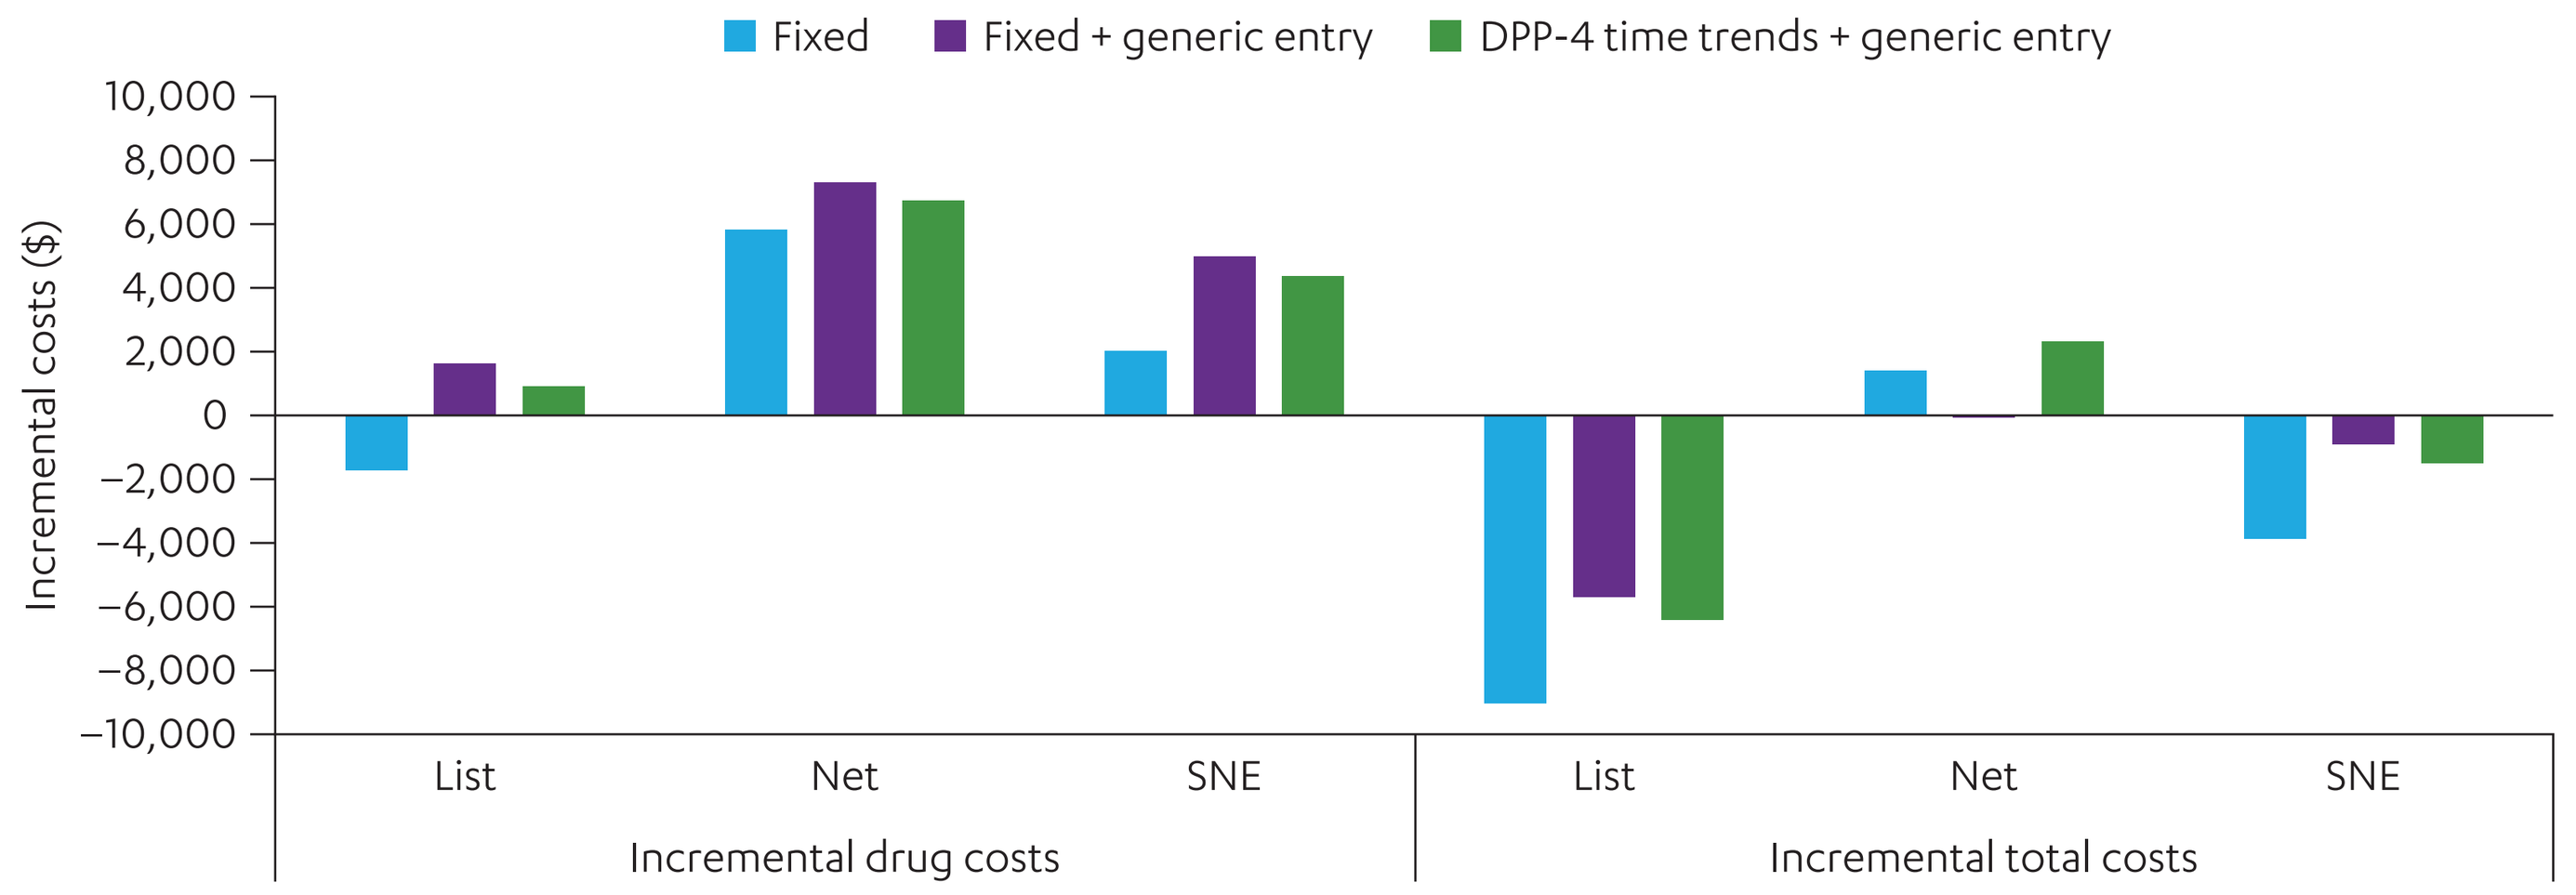
Fixed
Fixed + generic entry
DPP-4 time trends + generic entry
Incremental costs ($)
List
Net
SNE
Incremental drug costs
Incremental total costs

DPP-4, dipeptidyl peptidase-4; SNE, systemwide net expenditure.

DISCUSSION

- Value assessments are often informed by analyses using prices that ignore future price dynamics[5] and/or use price metrics that misstate true transaction prices,[2] despite best practice recommendations[3]
- Forecasted price trajectories were used to show how a researcher can model future branded and unbranded price competition. Not surprisingly, assumptions regarding the impacts of the entrance of generics into the market were a key value driver
- To our knowledge, this is the first study to consider the impact of price metrics and price dynamics simultaneously. Additionally, this study may be the first to consider the implications of branded price competition in the US setting, as well as SNE as an alternative to list and net prices in comparative economic research
- The limitations of using list prices to represent actual transaction prices are well known. Net prices are occasionally used but they exclude the growing share of costs captured by drug market intermediaries. SNE can better reflect the societal transaction price, and attempts should be made to introduce this into economic analysis
- The limitations of fixing list prices at baseline are well known, but we have demonstrated that analysts can create scenarios to model price changes
 - Economists have recognized the importance of modeling the impact of generic entry,[4,5,8] which is feasible in many cases (as generic entry is reasonably predictable)
 - Branded price competition has received extremely limited attention.[4] Future prices can be considered like other uncertain factors that are routinely modeled (e.g., durability of treatment effects, treatment switching patterns). We used a simple approach based only on actual price data that would have been available to a researcher at launch. More research into the determinants of price dynamics can improve on this approach

CONCLUSIONS

- Our results suggest that both the choice of price metric and assumptions about future price trajectories can have important impacts on estimates of value
- Using the conventional assumption of fixed list price systematically led to higher drug cost estimates compared to using both net prices and SNEs. These 2 price metrics are more important to consider than list price for many stakeholders in the US setting
- Ignoring market events (especially reasonably predictable ones) ensures that decision-makers will be misinformed

REFERENCES

1. IMS Institute for Healthcare Informatics. Price declines after branded medicines lose exclusivity in the U.S. Accessed March 21, 2023. https://www.iqvia.com/-/media/iqvia/pdfs/institute-reports/price-declines-after-branded-medicines-lose-exclusivity-in-the-us.pdf. 2. Van Nuys K, et al. *JAMA Health Forum*. 2021;2(11):e213409. 3. Hay JW, et al. *Value Health*. 2010;13(1):3-7. 4. Schöttler MH, et al. *Value Health*. 2023;26(1):91-98. 5. Neumann PJ, et al. *Value Health*. 2022;25(1):59-68. 6. Berndt ER, Aitken ML. *Int J Econ Bus*. 2011;18(2):177-201. 7. Nguyen NX, et al. *Appl Health Econ Health Policy*. 2022;20(2):243-253. 8. Cohen JT. *Value Health*. 2022;26(3):344-350. 9. Neal B, et al. *N Engl J Med*. 2017;377(7):644-657. 10. Willis M, et al. *Pharmacoeconomics*. 2017;35(3):375-396. 11. Willis M, et al. *Pharmacoeconomics*. 2020;38(9):953-969. 12. Neslusan C, et al. *Diabetes Ther*. 2018;9(2):565-581. 13. Schernthaner G, et al. *Diabetes Care*. 2013;36(9):2508-2515. 14. Sabapathy S, et al. *J Popul Ther Clin Pharmacol*. 2016;23(2):e151-e168. 15. Perkovic V, et al. *N Engl J Med*. 2019;380(24):2295-2306. 16. Willis M, et al. *Value Health*. 2017;20(3):357-371. 17. Ward A, et al. *J Med Econ*. 2014;17(3):176-183. 18. Hoerger TJ, et al. *Am J Kidney Dis*. 2010;55(3):463-473. 19. Pititto L, et al. Poster presented at: International Society for Pharmacoeconomics and Outcomes Research (ISPOR) 5th Latin America Conference; September 6-8, 2015; Santiago, Chile. 20. Thompson G, et al. Poster presented at: European Association for the Study of Diabetes (EASD) 50th Annual Meeting; September 15-19, 2014; Vienna, Austria.

DISCLOSURES

MW and AN are employees of The Swedish Institute for Health Economics, which has provided consulting services for Janssen Scientific Affairs, LLC. CN is an employee of Janssen Scientific Affairs, LLC.

ACKNOWLEDGMENTS

This study was sponsored by Janssen Scientific Affairs, LLC. Editorial assistance was provided by Alaina Mitsch, PhD, of Lumanity Communications Inc., and was funded by Janssen Scientific Affairs, LLC.

Presented at the Professional Society for Health Economics and Outcomes Research (ISPOR) Annual Meeting; May 7-10, 2023; Boston, MA.

This study was sponsored by Janssen Scientific Affairs, LLC.