HbA1c Reduction with Digital Health Devices in Type 2 Diabetes: A Meta-Analysis of Randomized Controlled Trials

Felix Lee[1], Edward Han-Burgess[2], Adee Kennedy[1], Paul Serafini[3], Mir-Masoud Pourrahmat[3], Boris Breznen[3]

[1]Sanofi, General Medicines, Bridgewater, NJ, United States of America, [2]Sanofi, General Medicines, Cambridge, MA, United States of America, [3]Evidinno Outcomes Research Inc., Evidence Synthesis, Vancouver, BC, Canada

Poster MT2

ISPOR 2023
May 7–10, 2023,
Boston, MA, USA

INTRODUCTION

- Type 2 diabetes mellitus (T2DM) is a metabolic disorder characterized by chronic hyperglycemia due to impaired insulin production and secretion.
- As of 2021, 537 million adults are living with diabetes, and this number is projected to increase to 783 million by 2045.[1]
- Many patients struggle to achieve glycemic control, putting them at risk of microvascular (e.g., retinopathy, nephropathy) and macrovascular complications (e.g., coronary heart disease, cardiomyopathy).[2]
- Glucose monitoring devices, such as self-measured blood glucose (SMBG) and continuous glucose monitoring (CGM), can help patients to achieve and maintain glycemic control.[2]
- In addition, evidence shows that patients need assistance and coaching for building awareness of their daily health-related behaviors.[3] This helps in bridging the gap between glucose awareness and behavioral change.

OBJECTIVE

To compare the efficacy of digital T2DM interventions, defined as SMBG or CGM combined with a coaching component, in reducing HbA1c compared to usual care.

METHODS

Systematic Literature Review

- A systematic review was conducted using standard methodologies from Cochrane. Details have been previously described,[4] briefly:
 - MEDLINE®, Embase, and Cochrane Central Register of Controlled Trials (CENTRAL) were searched from database inception to April 5, 2022. Searches were limited to the English language.
 - Included articles were comparative observational and clinical trials on adults (>18 years old) with T2DM who received a digital intervention (containing both human coaching and digital glucose monitoring components) or usual care.
 - The primary outcome of interest was change in HbA1c.

Meta-Analysis

- Randomized controlled trials (RCTs) were eligible for the meta-analysis (MA).
- If a study had more than one control or intervention arm, then the arms were pooled by taking the sample size-weighted average of mean change in HbA1c, and if a study reported HbA1c at more than one timepoint then the final measurement was used.
- MAs were conducted using the metafor package (v3.0-2)[5] for R (v4.1.1),[6] using the random effects model (restricted maximum likelihood method).
- The primary analysis was restricted to studies that reported lab measured HbA1c.
- In a secondary analysis, meta-regression was performed with intensity of coaching in the digital intervention (high, medium, or low) as a categorical covariate. Intensity was defined as follows:
 - **High intensity:** Patient data is uploaded automatically and regularly. Communication with dedicated staff occurs at least once per week, with personalized encouragement/goal-setting and education about the disease, behavioral strategies, and psychological coping.
 - **Medium intensity:** Patient data is uploaded manually. Communication is initiated ad-hoc by staff and includes non-behavioral advice and education on the disease and the digital device.
 - **Low-intensity:** Data sharing is limited. Communication is delayed, feedback is generic, and there is no education component.
- We also conducted sensitivity analyses (1) including studies which did not measure HbA1c in a lab, (2) excluding studies which were judged to have a high risk of bias according to the Cochrane risk of bias tool,[4] and (3) excluding studies which used CGM.
- We evaluated heterogeneity using tau, the Cochran's Q test, and the I^2 statistic.

RESULTS

Study Selection

- Of 6,288 records screened, 23 RCTs were eligible for the MA (See **Figure 1** for the PRISMA diagram and **Table 1** for list of included studies).
- Across the included studies, the median age was 55.6 years (range: 47.2 to 63.1 years), and the median proportion of female patients was 50.7% (29.0% to 72.6%). At baseline, the median HbA1c was 8.5% (7.0% to 10.9%), and the median BMI was 31.5 kg/m^2 (24.0 to 40.8 kg/m^2).
- Three studies did not measure HbA1c in a lab, and so were excluded from the primary analysis but included in a sensitivity analysis: Guo (2021), Pimazoni-Netto (2011), and Welch (2015).[7-9]
- Two studies used CGM and were excluded in a sensitivity analysis: Allen (2011) and Lee (2019).[10,11]
- Four studies were judged high risk of bias and were excluded in a sensitivity analysis: Allen (2011), Azelton (2021), and Kim (2003) because they did not employ double-blinding,[10,12,13] and Jeong (2018) because it did not adequately report how HbA1c was collected.[14]

Meta-Analysis

Primary Analysis

- The primary analysis estimated -0.31% (95% confidence interval [CI]: -0.45, -0.16; $p = 2.38 \times 10^{-5}$) greater reduction with a digital intervention compared to usual care (**Figure 2**).
- Heterogeneity was statistically significant (Q = 57.64, df = 19, $p = 9.09 \times 10^{-6}$), with an estimated τ of 0.21 (95% CI: 0.12, 0.61) and I^2 of 67.54% (95% CI: 41.2, 94.48).

Meta-Regression Analysis

- When digital intervention intensity was included as a categorical covariate, the following meta-regression equation was estimated:

$$\widehat{\mathrm{MD}} = -0.43 + 0.18(\text{intensity=low}) + 0.20(\text{intensity=medium}),$$

 where $\widehat{\mathrm{MD}}$ is the predicted mean difference (MD) of change in HbA1c, "intensity=low" is 1 if the digital intervention being predicted is low intensity and 0 otherwise, and "intensity=medium" is 1 if the digital intervention being predicted is medium intensity and 0 otherwise.
- This predicts an MD of -0.43% (95% CI: -0.74, -0.11; p = 0.0084) for high intensity interventions, -0.22% (95% CI: -0.38, -0.06; p = 0.0055) for medium intensity interventions, and -0.25% (95% CI: -0.57, 0.07; p = 0.13) for low intensity interventions (**Figure 1**).
- The low (p = 0.44) and medium (p = 0.26) intensity coefficients were not statistically significant.
- Heterogeneity was statistically significant (Q = 43.60, df = 19, p = 0.000393), with an estimated τ of 0.18 (95% CI: 0.11, 0.70) and I^2 of 54.45% (95% CI: 32.71, 94.90).

Figure 1: PRISMA Diagram

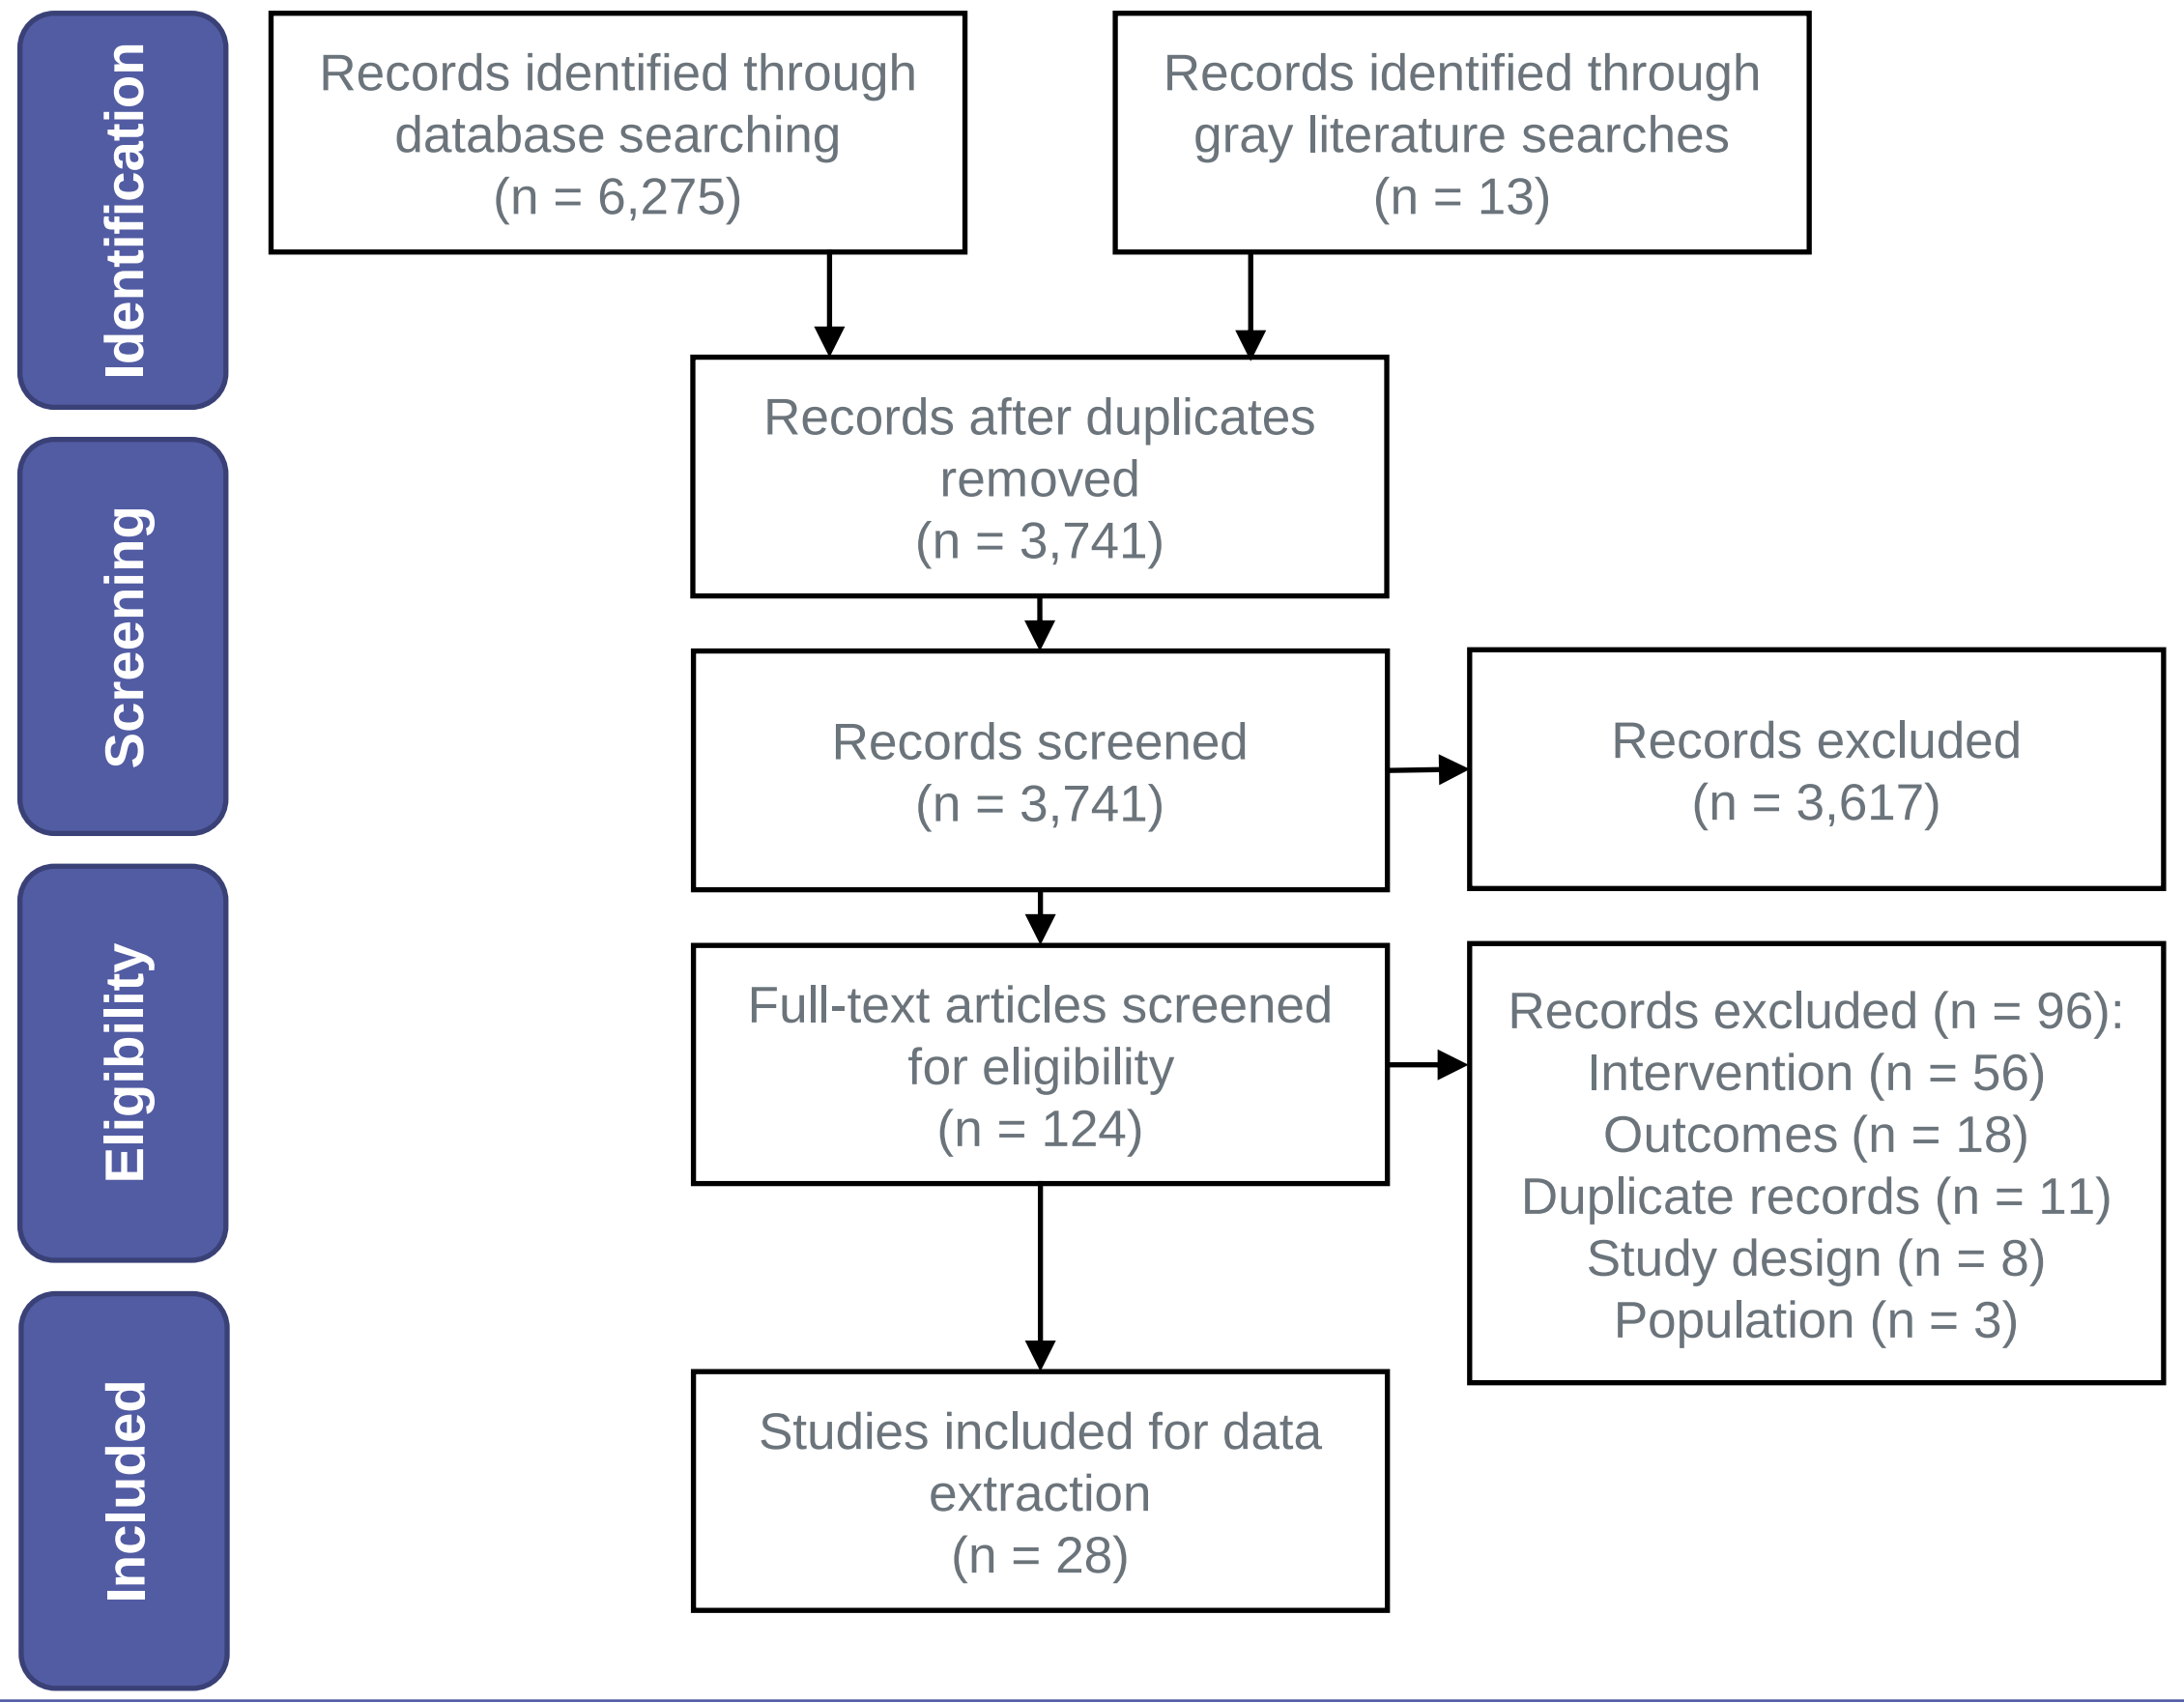
Identification
Screening
Eligibility
Included
Records identified through database searching (n = 6,275)
Records identified through gray literature searches (n = 13)
Records after duplicates removed (n = 3,741)
Records screened (n = 3,741)
Records excluded (n = 3,617)
Full-text articles screened for eligibility (n = 124)
Records excluded (n = 96): Intervention (n = 56) Outcomes (n = 18) Duplicate records (n = 11) Study design (n = 8) Population (n = 3)
Studies included for data extraction (n = 28)

Table 1: Included Studies

Study	Continent	Sample size	Follow-up (months)
Allen (2011)[10]	North America	29	3
Amante (2021)[15]	North America	119	12
Azelton (2021)[12]	North America	45	3
Cox (2021)[16]	North America	172	13
Guo (2021)[7]	Asia	68	1
Hee-Sung (2007)[17]	Asia	60	3
Hsu (2016)[18]	North America	40	3
Jeong (2018)[14]	Asia	338	6
Ji (2019)[19]	Asia	100	6
Kim (2003)[13]	Asia	50	3
Kirk (2009)[20]	Europe	134	12
Lee (2018)[21]	Asia	148	12
Lee (2020)[22]	Asia	240	12
Lee (2019)[11]	Asia	63	6
Nagrebetsky (2013)[23]	Europe	17	6
Odnoletkova (2016)[24]	Europe	574	18
Parsons (2019)[25]	Europe	446	12
Pimazoni-Netto (2011)[8]	North America	63	3
Quinn (2011)[26]	North America	213	12
Turnin (2021[27]	Europe	282	12
Wayne (2015)[28]	North America	138	6
Welch (2015)[9]	North America	399	6
Yang (2020)[29]	Asia	247	3

Note: Follow-up is in months. N/R – Not Reported.

Figure 2: Primary and Meta-Regression Analysis Results

Study	Intensity	Weight (%)	MD (95% CI)
Allen (2011)	High	0.9	-0.20 (-1.53, 1.13)
Amante (2021)	High	6.4	-0.37 (-0.74, 0.00)
Azelton (2021)	High	1.8	-0.70 (-1.60, 0.20)
Cox (2021)	Medium	4.5	-0.30 (-0.79, 0.19)
Hee-Sung (2007)	Medium	1.5	* -1.45 (-2.44, -0.46)
Hsu (2016)	Medium	1.2	* -1.20 (-2.30, -0.10)
Jeong (2018)	Medium	8.4	-0.07 (-0.35, 0.20)
Ji (2019)	Low	2.2	* -1.17 (-1.96, -0.38)
Kim (2003)	Medium	0.7	* -1.80 (-3.28, -0.32)
Kirk (2009)	Low	3.1	-0.10 (-0.73, 0.54)
Lee (2018)	Medium	6.2	0.10 (-0.28, 0.48)
Lee (2020)	Medium	13.4	-0.03 (-0.08, 0.02)
Lee (2019)	Medium	3.5	-0.41 (-1.00, 0.18)
Nagrebetsky (2013)	Medium	0.9	-0.40 (-1.71, 0.91)
Odnoletkova (2016)	Low	10.6	-0.10 (-0.29, 0.09)
Parsons (2019)	Medium	7.1	-0.12 (-0.45, 0.21)
Quinn (2011)	High	3.0	* -0.99 (-1.65, -0.34)
Turnin (2021)	Medium	9.7	-0.20 (-0.42, 0.02)
Wayne (2015)	High	4.5	-0.06 (-0.56, 0.44)
Yang (2020)	Medium	10.3	* -0.35 (-0.55, -0.15)
Summary	**High**	**100.0**	*** -0.43 (-0.74, -0.11)**
Summary	**Medium**	**100.0**	*** -0.22 (-0.38, -0.06)**
Summary	**Low**	**100.0**	**-0.25 (-0.57, 0.07)**
Summary	**Total**	**100.0**	*** -0.31 (-0.45, -0.16)**

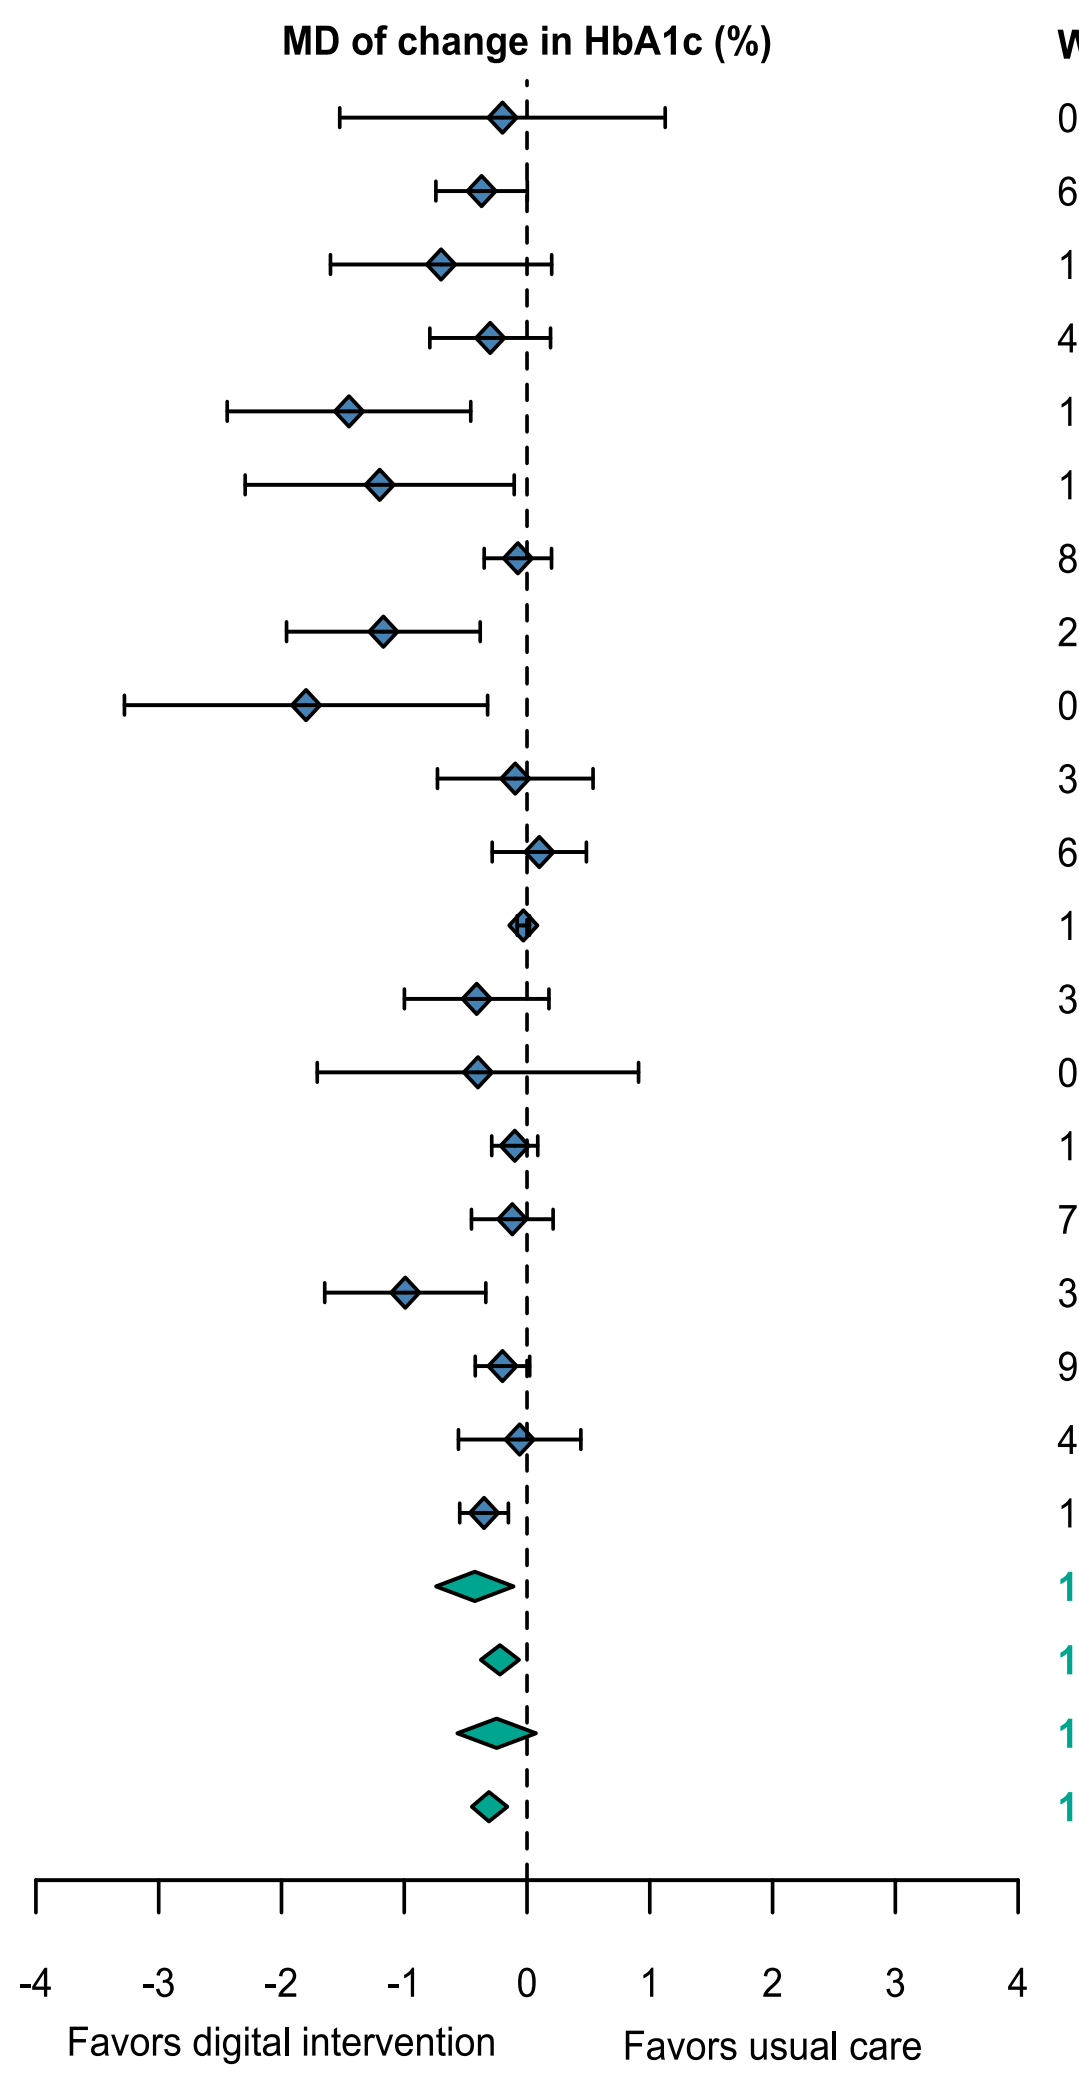
MD of change in HbA1c (%)
-4
-3
-2
-1
0
1
2
3
4
Favors digital intervention
Favors usual care

Note: The "Total" summary row is from the primary analysis, and the "High", "Medium", and "Low" summary rows are from the meta-regression analysis. * – A statistically significant comparison (p ≤ 0.05), CI – Confidence Interval, MD – Mean Difference.

RESULTS (Continued)

Sensitivity Analyses

- When studies with non-lab measured HbA1c were included, the resulting MD was more favorable to the digital intervention (MD: -0.40%; 0.95% CI: -0.56, -0.24; $p = 7.64 \times 10^{-7}$).
- When CGM studies (MD: -0.31%; 0.95% CI: -0.47, -0.16; $p = 6.16 \times 10^{-5}$) and studies with high risk of bias (MD: -0.31%; 0.95% CI: -0.46, -0.15; $p = 8.41 \times 10^{-5}$) were excluded, the point estimate was the same as the primary analysis, but the CI was slightly wider.

CONCLUSIONS

- **This meta-analysis found significantly greater HbA1c reduction in T2DM patients on digital interventions compared to usual care.**
 - Including studies with non-lab measured HbA1c led to a more favorable result for the digital intervention.
 - Excluding CGM studies or those with high risk of bias led to similar results as compared with the primary analysis.
- Results support digital interventions as an effective addition to usual care.
- The association between digital intervention intensity and HbA1c reduction was not statistically significant, and further research is warranted to understand optimal intensity for digital interventions.

REFERENCES

1. International Diabetes Federation. https://diabetesatlas.org/. [Accessed April 11, 2023].
2. Bode B, et al. Leveraging advances in diabetes technologies in primary care: a narrative review. *Annals of Medicine*. 2021;53(1):805-816.
3. Fundoiano-Hershcovitz Y, et al. Role of Digital Engagement in Diabetes Care Beyond Measurement: Retrospective Cohort Study. *JMIR Diabetes*. 2021;6(1):e24030.
4. Lee FC, et al. Digital interventions for self-management in type 2 diabetes mellitus (T2DM): a systematic literature review. *Diabetes Technology & Therapeutics*. 2023;25(S2):A-255.
5. Viechtbauer W. Conducting Meta-Analyses in R with the metafor Package. *Journal of Statistical Software*. 2010;36(3):1-48.
6. Team RC. R: A language and environment for statistical computing. In: Vienna, Austria; 2013.
7. Guo M, et al. Effectiveness of mHealth management with an implantable glucose sensor and a mobile application among Chinese adults with type 2 diabetes. *Journal of Telemedicine and Telecare*. 2021;0(0).
8. Pimazoni-Netto A, et al. Rapid improvement of glycemic control in type 2 diabetes using weekly intensive multifactorial interventions: Structured glucose monitoring, patient education, and adjustment of therapy - a randomized controlled trial. *Diabetes Technology & Therapeutics*. 2011;13(10):997-1004.
9. Welch G, et al. An internet-based diabetes management platform improves team care and outcomes in an urban Latino population. *Diabetes Care*. 2015;38(4):561-567.
10. Allen N, et al. A continuous glucose monitoring and problem-solving intervention to change physical activity behavior in women with type 2 diabetes: A pilot study. *Diabetes Technology & Therapeutics*. 2011;13(11):1091-1099.
11. Lee SK, et al. Effect of diabetes education through pattern management on self-care and self-efficacy in patients with type 2 diabetes. *International Journal of Environmental Research and Public Health*. 2019;16(18).
12. Azelton KR, et al. Digital Health Coaching for Type 2 Diabetes: Randomized Controlled Trial of Healthy at Home. *Frontiers in Digital Health*. 2021;3;172.
13. Kim HS, et al. Adherence to diabetes control recommendations: impact of nurse telephone calls. *Journal of Advanced Nursing*. 2003;1(3):256-261.
14. Jeong JY, et al. Smart Care Based on Telemonitoring and Telemedicine for Type 2 Diabetes Care: multi-Center Randomized Controlled Trial. *Telemedicine Journal and e-Health*. 2018;24(8):604-613.
15. Amante DJ, et al. Evaluation of a Diabetes Remote Monitoring Program Facilitated by Connected Glucose Meters for Patients With Poorly Controlled Type 2 Diabetes: Randomized Crossover Trial. *JMIR Diabetes*. 2021;1(1):e25574.
16. Cox DJ, et al. Long-term follow-up of a randomized clinical trial comparing glycemic excursion minimization (GEM) to weight loss (WL) in the management of type 2 diabetes. *BMJ Open Diabetes Research & Care*. 2021;9(2).
17. Hee-Sung K. Impact of Web-based nurse's education on glycosylated haemoglobin in type 2 diabetic patients. *Journal of Clinical Nursing*. 2007;16(7):1361-1366.
18. Hsu WC, et al. Utilization of a Cloud-Based Diabetes Management Program for Insulin Initiation and Titration Enables Collaborative Decision Making Between Healthcare Providers and Patients. *Diabetes Technology & Therapeutics*. 2016;18(2):59-67.
19. Ji H, et al. Effect of simulation education and case management on glycemic control in type 2 diabetes. *Diabetes/Metabolism Research and Reviews*. 2019;35(3):e3112.
20. Kirk A, et al. A randomized trial investigating the 12-month changes in physical activity and health outcomes following a physical activity consultation delivered by a person or in written form in Type 2 diabetes: time2Act. *Diabetic Medicine*. 2009;26(3):293-301.
21. Lee DY, et al. The effectiveness, reproducibility, and durability of tailored mobile coaching on diabetes management in policyholders: a randomized, controlled, open-label study. *Scientific Reports*. 2018;8(1):3642.
22. Lee JY, et al. Telemonitoring and Team-Based Management of Glycemic Control on People with Type 2 Diabetes: a Cluster-Randomized Controlled Trial. *Journal of General Internal Medicine*. 2020;35(1):87-94.
23. Nagrebetsky A, et al. Stepwise self-titration of oral glucose-lowering medication using a mobile telephone-based telehealth platform in type 2 diabetes: a feasibility trial in primary care. *Journal of Diabetes Science and Technology*. 2013;7(1):123-134.
24. Odnoletkova I, et al. Optimizing diabetes control in people with Type 2 diabetes through nurse-led telecoaching. *Diabetic Medicine*. 2016;33(6):777-785.
25. Parsons SN, et al. Effect of structured self-monitoring of blood glucose, with and without additional TeleCare support, on overall glycaemic control in non-insulin treated Type 2 diabetes: the SMBG Study, a 12-month randomized controlled trial. *Diabetic Medicine*. 2019;36(5):578-590.
26. Quinn CC, et al. Cluster-randomized trial of a mobile phone personalized behavioral intervention for blood glucose control. *Diabetes Care*. 2011;34(9):1934-1942.
27. Turnin MC, et al. Impact of a Remote Monitoring Programme Including Lifestyle Education Software in Type 2 Diabetes: results of the Educ@dom Randomised Multicentre Study. *Diabetes Therapy*. 2021;12(7):2059-2075.
28. Wayne N, et al. Health Coaching Reduces HbA1c in Type 2 Diabetic Patients From a Lower-Socioeconomic Status Community: A Randomized Controlled Trial. *Journal Of Medical Internet Research*. 2015;17(10):e224.
29. Yang Y, et al. Effect of a Mobile Phone-Based Glucose-Monitoring and Feedback System for Type 2 Diabetes Management in Multiple Primary Care Clinic Settings: Cluster Randomized Controlled Trial. *JMIR mHealth and uHealth*. 2020;8(2):e16266.

DISCLOSURES

This study was funded by Sanofi and conducted by Evidinno Outcomes Research Inc. Felix Lee, Edward Han-Burgess, and Adee Kennedy are employees of Sanofi and may hold shares/stock in Sanofi. Paul Serafini, Mir-Masoud Pourrahmat, and Boris Breznen report employment with Evidinno Outcomes Research Inc. Authors report no other conflicts of interest.

FUNDING

The study was sponsored by Sanofi. Medical writing and poster preparation support was provided by Mariana Machado from Evidinno Outcomes Research Inc. (Vancouver, Canada), funded by Sanofi.