Characterization of OCS Use in Patients Starting Biologics for Asthma in the United States

Poster No. CO34

Authors:
Harvey J[1], Zhang S[1], Mu G[1], Valliant SN[1], Alfonso-Cristancho R[1]

Affiliations:
[1]*Value Evidence Outcomes, GSK, Upper Providence, PA, USA*

Introduction

Patients with asthma may require OCS to help control their symptoms.[1]

A main aim of severe asthma management is to reduce and eliminate maintenance and burst use of OCS because of their associated adverse effects.[1,2]

Multiple biologics are approved for patients with asthma in the USA, with an OCS-sparing effect demonstrated across clinical and real-world studies.[3–12]

Aim

The aim of this analysis was to describe the burden of OCS use in patients with asthma who were initiating a biologic therapy in the USA.

Year	Biologic	Indication	Age	Target
2003	Omalizumab (O)	Severe allergic asthma	≥6 years of age	anti-IgE
2015	Mepolizumab (M)	Severe allergic asthma	≥6 years of age	anti-IL-5
2016	Reslizumab (R)	Severe eosinophilic asthma	≥18 years of age	anti-IL-5
2017	Benralizumab (B)	Severe eosinophilic asthma	≥12 years of age	anti-IL-5Rα
2018	Dupilumab (D)	Severe asthma with T2 inflammation	≥6 years of age	anti-IL-4Rα

Study design

- GSK ID: 219356
- Retrospective cohort study
- Optum's de-identified Clinformatics® Data Mart Database

Patients with asthma

- Initiated biologic therapy during the study period
- Continuous enrollment with pharmacy benefits during the baseline period
- ≥1 diagnosis of asthma (any position) during baseline period
- ≥12 years of age at index

- Patients with a biologic prescription from a "dermatology" provider*

Jan

Baseline period: 12 months prior to Index

Index date: biologic therapy intiation

Study period

2022

Mar

Outcomes

Differences in baseline OCS use in patients:

With mOCS† use (n=1968) | Without mOCS† use (n=7305)

- **Total OCS‡ use**
 Daily dose§; prescription count; days supplied
- **mOCS use**

*Due to the low probability of dermatologists prescribing a biologic for asthma as the primary diagnosis; some patients may have still been included with a primary reason of prescribing biologics for a condition other than asthma; †mOCS was defined as a prescription >28 days; ‡inclusive of mOCS and OCS bursts defined as 3–28 days supply of ≥20 mg prednisone equivalent OCS dose in patients with an asthma diagnosis within 2 weeks of the biologic prescription date; §daily OCS dose was calculated by averaging the dose across all days assessed, including the days without OCS.

Results

9273 patients

21% had mOCS use

Patient demographics

Scan me to see baseline demographics stratified by each biologic

	With mOCS use	Without mOCS use
Age, years, %		
12–64	59	69
≥65	41	31
Female, %	62	65
Health care coverage, %		
Commercial	47	61
Medicare	53	39
Patients with exacerbations*, %		
≥1	64	54
≥4	14	8
Asthma duration, years, mean (SD)	4.4 (4.1)	4.7 (4.5)
CCI, mean (SD)	2.0 (1.5)	1.7 (1.3)

Fewer patients with mOCS use were ≤64 years of age

More patients with mOCS use had Medicare coverage

Patients with mOCS use had a greater CCI

Comorbidities†, %	With mOCS use	Without mOCS use
Infection: respiratory	65 ◎ ↑	56 ◎
Allergic rhinitis	60 ◎ ↓	70 ◎
COPD	50 ↑	29
Chronic rhinosinusitis	33	30
Infection: other viral	17	17
Atopic dermatitis	14 ↓	27
Chronic rhinitis	14	14
CRSwNP	13	16
Chronic idiopathic urticaria	7	12

◎ Common comorbidities; Comorbidity prevalence (with vs without mOCS use): ↑ Higher ↓ Lower

*Index specialty of dermatologist was excluded; †comorbidities with >10% prevalence for at least one biologic are shown.

Baseline daily total OCS dose, mg (SD)

Daily total OCS dose was higher in patients with mOCS use than patients without OCS use; patients without mOCS use still had ~2 mg/day exposure (to OCS bursts).

◎ Highest daily OCS dose

Baseline total OCS prescription count, mean (SD)

A higher number of prescriptions was observed in patients with mOCS use; however, even patients without mOCS use continued to have OCS prescriptions for OCS bursts.

◎ Highest prescription count

Baseline total OCS prescriptions, days supplied, mean (SD)

The mean days supplied for total OCS prescriptions was highest in patients with mOCS use, although patients without mOCS use still averaged >8 days supplied (for OCS bursts).

◎ Highest prescription count; △ Smallest difference between subgroups

	n	Daily dose, with mOCS	Daily dose, without mOCS	Prescription count, with mOCS	Prescription count, without mOCS	Days supplied, with mOCS	Days supplied, without mOCS
Total	(N=9273)	7.4 (5.6)	1.8 (2.3)	7.0 (4.3)	2.8 (3.0)	24.1 (21.1)	8.8 (4.8)
O	(n=2805)	6.8 (5.2)	1.5 (1.9)	6.6 (4.0)	2.4 (2.6)	24.1 (21.5)	8.1 (4.5)
M	(n=1642)	◎ 8.1 (5.3)	◎ 2.3 (2.8)	◎ 7.4 (4.1)	◎ 3.3 (3.6)	◎ 25.1 (22.4)	9.0 (4.9)
R	(n=80)	◎ 9.5 (7.8)	◎ 2.2 (2.7)	7.2 (5.2)	3.0 (3.0)	△ 22.1 (19.6)	△ 9.5 (5.1)
B	(n=1728)	7.2 (5.2)	2.1 (2.4)	7.0 (4.0)	3.1 (3.1)	24.4 (21.3)	8.4 (4.6)
D	(n=3018)	7.4 (6.2)	1.6 (2.3)	6.9 (4.2)	2.3 (2.5)	24.7 (20.9)	8.8 (4.9)

Baseline mOCS prescription count in patients with mOCS use*

	n	Prescription count, mean (SD)	Prescription category, %: Both mOCS and bursts*	Prescription category, %: mOCS only
Total	N=1968	3.4 (3.0)	64	36
O	n=494	3.2 (3.1)	64	36
M	n=535	◎ 3.6 (3.4)	69	31
R	n=31	3.1 (2.2)	61	39
B	n=421	3.5 (3.2)	66	34
D	n=487	◎ 3.6 (3.3)	56	44

◎ Had the greatest number of mOCS prescriptions

Most patients had prescriptions for mOCS and OCS bursts

*Patients without baseline mOCS use had a mean (SD) prescription count of 0.0 (0.0) across the biologics.

Conclusions

Our data highlight that, despite current guidance to minimize OCS use,[1] over 20% of patients with asthma were using mOCS before initiating a biologic, indicative of a substantial clinical and economic burden.[1,2,14]

- Even in patients without mOCS use, the lower exposure to OCS bursts reported in this analysis, has been associated with adverse effects and increased healthcare costs.[2,14]
- Patients initiating mepolizumab generally had the greatest baseline OCS use.

Together these data highlight that patients with asthma may benefit from timely intervention with biologics, given their OCS-sparing effect.[8–12]

Abbreviations

CCI, Charlson Comorbidity Index; COPD, chronic obstructive pulmonary disease; CRSwNP, chronic rhinosinusitis with nasal polyps; IgE, immunoglobulin E; IL, interleukin; mOCS, maintenance OCS; OCS, oral corticosteroid; SD, standard deviation; T2, type 2; USA, United States of America.

Disclosures

- This study was funded by GSK (GSK ID: 219356).
- **JH** and **SNV** are university staff members employed by GSK. **SZ**, **GM**, and **RA-C** are employees of GSK and hold stock/shares in GSK.
- Editorial support (in the form of writing assistance, including preparation of the draft poster under the direction and guidance of the authors, collating and incorporating authors' comments for each draft, assembling tables and figures, grammatical editing and referencing) was provided by Katie Crossland, PhD, at Fishawack Indicia Ltd, part of Fishawack Health, UK, and was funded by GSK.

References

1. Global Initiative for Asthma. Global strategy for asthma management and prevention 2022. Available from: https://ginasthma.org/wp-content/uploads/2022/07/GINA-Main-Report-2022-FINAL-22-07-01-WMS.pdf [last accessed March 11, 2023].
2. Bloechliger M, et al. *Respir Res* 2018;19(1):75.
3. AstraZeneca AB. Fasenra Prescribing Information. Updated 2022. Available from: https://www.accessdata.fda.gov/drugsatfda_docs/label/2017/761070s000lbl.pdf [last accessed March 11, 2023].
4. GSK. Nucala Prescribing Information. Updated 2022. Available from: https://www.accessdata.fda.gov/drugsatfda_docs/label/2022/761122s008,125526s019lbl.pdf [last accessed March 11, 2023].
5. Teva Pharmaceuticals. Cinqaero Prescribing Information. Updated 2016. Available from: https://www.accessdata.fda.gov/drugsatfda_docs/label/2016/761033lbl.pdf [last accessed March 11, 2023].
6. Sanofi Winthrop Industrie. Duxipent Prescribing Information. Updated 2017. Available from: https://www.accessdata.fda.gov/drugsatfda_docs/label/2022/761055s042lbl.pdf [last accessed March 11, 2023].
7. Novartis Europharm Limited. Xolair Prescribing Information. Updated 2016. Available from: https://www.accessdata.fda.gov/drugsatfda_docs/label/2016/103976s5225lbl.pdf [last accessed March 11, 2023].
8. Celeste M, et al. *J Allergy Clin Immunol Pract* 2022;10(5):1202–16.
9. Bousquet J, et al. *J Allergy Clin Immunol Pract* 2021;9(7):2702–14.
10. Pilette C, et al. *J Allergy Clin Immunol Pract* 2022;10(10):2646–56.
11. Charles D, et al. *Clin Exp Allergy* 2022;52(5):616–27.
12. Kavanagh JE, et al. *Chest* 2021;159(2):496–506.
13. Quan H, et al. *Med Care* 2005;43:1130–39.
14. Barry LE, et al. *Respir Res* 2017;18(1):129.

Scan me
QR code to access a downloadable version of this poster

Presented at ISPOR, Boston, MA, USA, May 7–10, 2023