Beyond conventional clinical value: estimating the impact of accelerated approvals on therapeutic bridging between innovative therapies

HPR6

William Wong,[1] Stacey Kowal,[1] Tu My To,[1] Achal Patel,[1] David Veenstra,[2,3] Louis Garrison,[2,4] Meng Li[5]

[1]Genentech, Inc., South San Francisco, CA, USA; [2]The CHOICE Institute, School of Pharmacy, University of Washington, Seattle, WA, USA; [3]Curta, Inc., Seattle, WA, USA; [4]VeriTech Corporation, Mercer Island, WA, USA; [5]Department of Health Services Research, University of Texas MD Anderson Cancer Center, Houston, TX, USA

INTRODUCTION

- Drugs that improve survival may create real option value (ROV) by providing patients with a bridge to future innovations.
- In the US market, the Accelerated Approval (AA) Program, a regulatory pathway that allows the Food and Drug Administration (FDA) to more expeditiously approve drugs that treat serious conditions with high unmet need based on a surrogate endpoint,[1] has contributed to these advances in therapy options.
- Information on the net impact and value of the AA Program in oncology is needed to better inform the ongoing public discussion of its benefits and challenges.[2]

OBJECTIVE

- To study the impact of recent AAs in oncology by evaluating real-world data for treatments in three different tumors as case studies.

METHODS

- Data from the nationwide Flatiron Health electronic health record-derived, de-identified, longitudinal database were used to estimate patient-level outcomes for case studies in tumors with high levels of clinical innovation, and for which patients receiving recently approved products with extended survival could be eligible for future AA products within current survival estimates.
 - The Flatiron Health database is a longitudinal database, comprising de-identified patient-level structured and unstructured data, curated via technology-enabled abstraction.
- Metastatic melanoma, advanced bladder cancer and epidermal growth factor receptor positive (EGFR+) advanced non-small cell lung cancer (aNSCLC) were selected as case studies with the following criteria.
 - Tumor had an AA therapy option between 2011 (date of data availability) and 2021.
 - Cancer type had a newly approved 'initial drug' in a previous line of therapy to the AA drug indication.
 - Cancer type information was available to the research team from the Flatiron Health database, and there was a sufficient sample size to conduct the analysis (i.e. rare tumors were excluded).
- To estimate outcomes associated with the AA pathway, observed outcomes were compared with a counterfactual (hypothetical) scenario in which approval occurred at a later time through a traditional approval pathway.
 - Three counterfactual dates to simulate traditional FDA approval without AA were used to calculate outcomes: dates of 1) conference data presentation for the next innovation, 2) AA conversion to traditional approval and 3) average time from AA to full approval in oncology.
- Overall survival, stratified by type of follow-on therapy (next innovation, no treatment, standard of care [SoC]), was estimated using inverse probability of treatment weighted Kaplan–Meier curves.
- To assess proportions of patients receiving future AA drug(s) and survival outcomes, patients were followed up from their initiation of the newly approved drug(s) (referred to as 'initial drug' hereafter) in the three tumor types in the pre-AA period, defined as the period after the approval of the initial drug until the date of the AA of the future drug (**Table 1**).
 - The initial drugs examined were first-line ipilimumab (metastatic melanoma), post-platinum cancer immunotherapies (advanced bladder cancer), and first- and second-generation EGFR tyrosine kinase inhibitors (TKIs) (EGFR+ aNSCLC).

PLAIN LANGUAGE SUMMARY

What is this study about?

- The accelerated approval pathway of the US Food and Drug Administration provides patients with access to innovative drugs sooner than standard regulatory pathways. This study measured how many patients on currently approved cancer drugs received future cancer drugs because of the accelerated approval pathway and whether quicker access resulted in their living longer.

What were the results?

- The accelerated approval pathway led to more patients being able to receive future cancer drugs and therefore live longer.

What do the results of the study mean?

- The accelerated approval pathway is important because it improves outcomes of current cancer drugs by giving patients additional treatments, which allows patients to live longer to benefit from future treatments. Policymakers should consider this when thinking about making changes to the accelerated approval pathway.

RESULTS

Mean survival gains with AA drugs in the real-world database

- The difference in mean survival for patients treated with the initial drug and subsequently treated with the AA drug compared with SoC/no treatment was highest for the cancer immunotherapies in second-line and beyond metastatic melanoma (31.3 months), followed by EGFR+ aNSCLC (24.3 months) and advanced bladder cancer (13.6 months).
- Among patients treated with the initial drug, the average additional survival gain due to faster access to future drugs via the AA pathway was 7.0 months in metastatic melanoma, 5.2 months in EGFR+ aNSCLC and 1.2 months in advanced bladder cancer.

Table 1. Included drugs and key approval dates

	Metastatic melanoma	Advanced bladder cancer	EGFR+ aNSCLC
New drug/indication	1L ipilimumab	2L+ post-platinum immunotherapy[d]	EGFR+ erlotinib, gefitinib or afatinib
Traditional approval date	March 28, 2011	May 18, 2016 (atezolizumab)[b]	May 14, 2013 (erlotinib)[b]
Next innovation (AA product[s])	Immunotherapy (pembrolizumab and nivolumab)	Enfortumab vedotin-ejfv	Osimertinib
AA date	September 4, 2014 (pembrolizumab)[b]	December 18, 2019	November 13, 2015
Counterfactual AA dates			
Date of data presentation	April 19, 2015[c]	February 12, 2021[c]	December 6, 2016[c]
Date of actual full approval	December 18, 2015 (pembrolizumab)[b]	July 9, 2021	March 30, 2017
Average oncology conversion time[a]	March 4, 2017	June 17, 2022	May 13, 2018
Assumed delay in access to AA drug, months			
Date of data presentation	7.6	14.1	13.0
Date of conversion to full approval	15.7	19.0	16.8
Average oncology conversion time	30.4	30.4	30.4

[a]Average time for an oncology product approved through the AA pathway to convert to traditional approval, in months, based on oncology products with completed confirmatory trials per Center for Drug Evaluation and Research data on accelerated approvals available at: https://www.fda.gov/drugs/nda-and-bla-approvals/accelerated-approvals. [b]For instances in which there is more than one drug, we used the drug with the earliest approval dates. [c]Pembrolizumab data presented at the AACR Annual Meeting (metastatic melanoma), enfortumab vedotin-ejfv data presented at the ASCO GU Cancers Symposium (advanced bladder cancer), osimertinib data published in *New England Journal of Medicine* and presented at the World Conference on Lung Cancer. [d]Includes atezolizumab, nivolumab, durvalumab, avelumab and pembrolizumab.
1L, first-line; 2L+, second-line and beyond; AA, accelerated approval; AACR, American Association for Cancer Research; aNSCLC, advanced non-small cell lung cancer; ASCO, American Society of Clinical Oncology; EGFR+, epidermal growth factor receptor positive; GU, genitourinary.

Table 2. Population outcomes for AA pathway compared with counterfactual scenarios in which approval occurred at a later time through a traditional approval pathway

	Metastatic melanoma	Advanced bladder cancer	EGFR+ aNSCLC
Proportion surviving and receiving AA drug			
Total number of initial drug-treated patients	4394	21 488	20 317
Proportion surviving to AA date for future drug, %	40	31	62
Number of patients receiving future AA drug	745	1698	4233
Proportion surviving and treated with future AA drug among all initial drug patients, %	17	8	21
Average survival gain among all new drug patients, months	5.3	1.3	5.1
Primary analysis: traditional approval = date of AA conversion			
Number of additional patients receiving future AA drug owing to AA pathway	523	1247	2430
Average survival gain among all initial drug patients due to the AA pathway, months[a]	3.73	0.94	2.89
Survival gain among all initial drug patients attributable to AA pathway, %	70	73	57
Ratio of survival gain with AA pathway to traditional pathway	3.4	3.8	2.3
Population LYs gained with AA pathway	1367	1702	4930
Sensitivity analysis 1: shorter time to traditional approval			
Number of additional patients receiving future AA drug owing to AA pathway	355	1034	1942
Average survival gain among all initial drug patients due to the AA pathway, months[a]	2.53	0.78	2.31
Survival gain among all initial drug patients attributable to AA pathway, %	48	61	46
Ratio of survival gain with AA pathway to traditional pathway	1.9	2.6	1.8
Population LYs gained with AA pathway	927	1411	3934
Sensitivity analysis 2: longer time to traditional approval			
Number of additional patients receiving future AA drug owing to AA pathway	678	1571	3473
Average survival gain among all initial drug patients due to the AA pathway, months[a]	4.84	1.19	4.15
Survival gain among all initial drug patients attributable to AA pathway, %	91	92	82
Ratio of survival gain with AA pathway to traditional pathway	11.1	13.3	5.6
Population LYs gained with AA pathway	1772	2147	7075

[a]Estimate reflects the probability of receipt for the future AA drug based on the survival curve for the initial drug, and is therefore weighted (downwards) by the likelihood of surviving to the future AA drug.
AA, accelerated approval; aNSCLC, advanced non-small cell lung cancer; EGFR+, epidermal growth factor receptor positive; LY, life-year.

Mean survival gains attributable to the AA pathway in the US population

- The number of patients using the initial drug subsequently receiving the AA drug due to the AA pathway was 523/745 in metastatic melanoma, 1247/1698 in advanced bladder cancer and 2430/4233 in EGFR+ aNSCLC (**Table 2**).
- The number of patients receiving the future AA drug (due to the AA pathway) and the average additional survival gains due to the AA pathway was largest for patients who initiated their initial drug closest to the AA approval date (**Figure 1**).
 - An additional 5.3 months of survival (range: 0–13.2 months) for ipilimumab (metastatic melanoma) and 1.3 months (range: 0.3–3.0) for post-platinum cancer immunotherapies (advanced bladder cancer) were gained compared with the counterfactual scenario without AA.
 - Earlier access to next innovations via AA led to 355–678 more patients surviving to receive checkpoint inhibitors (metastatic melanoma) and 664–1571 more to receive enfortumab (advanced bladder cancer), resulting in 927–1772 life-years (LYs) gained (metastatic melanoma) and 1411–2147 LYs gained (advanced bladder cancer) at the population level.
 - Survival gains attributable to the AA pathway ranged from 46% to 61% and LYs gained ranged from 827 to 38 934.

Figure 1. Average additional survival (in months) per patient with and without AA pathway by proximity to the AA date

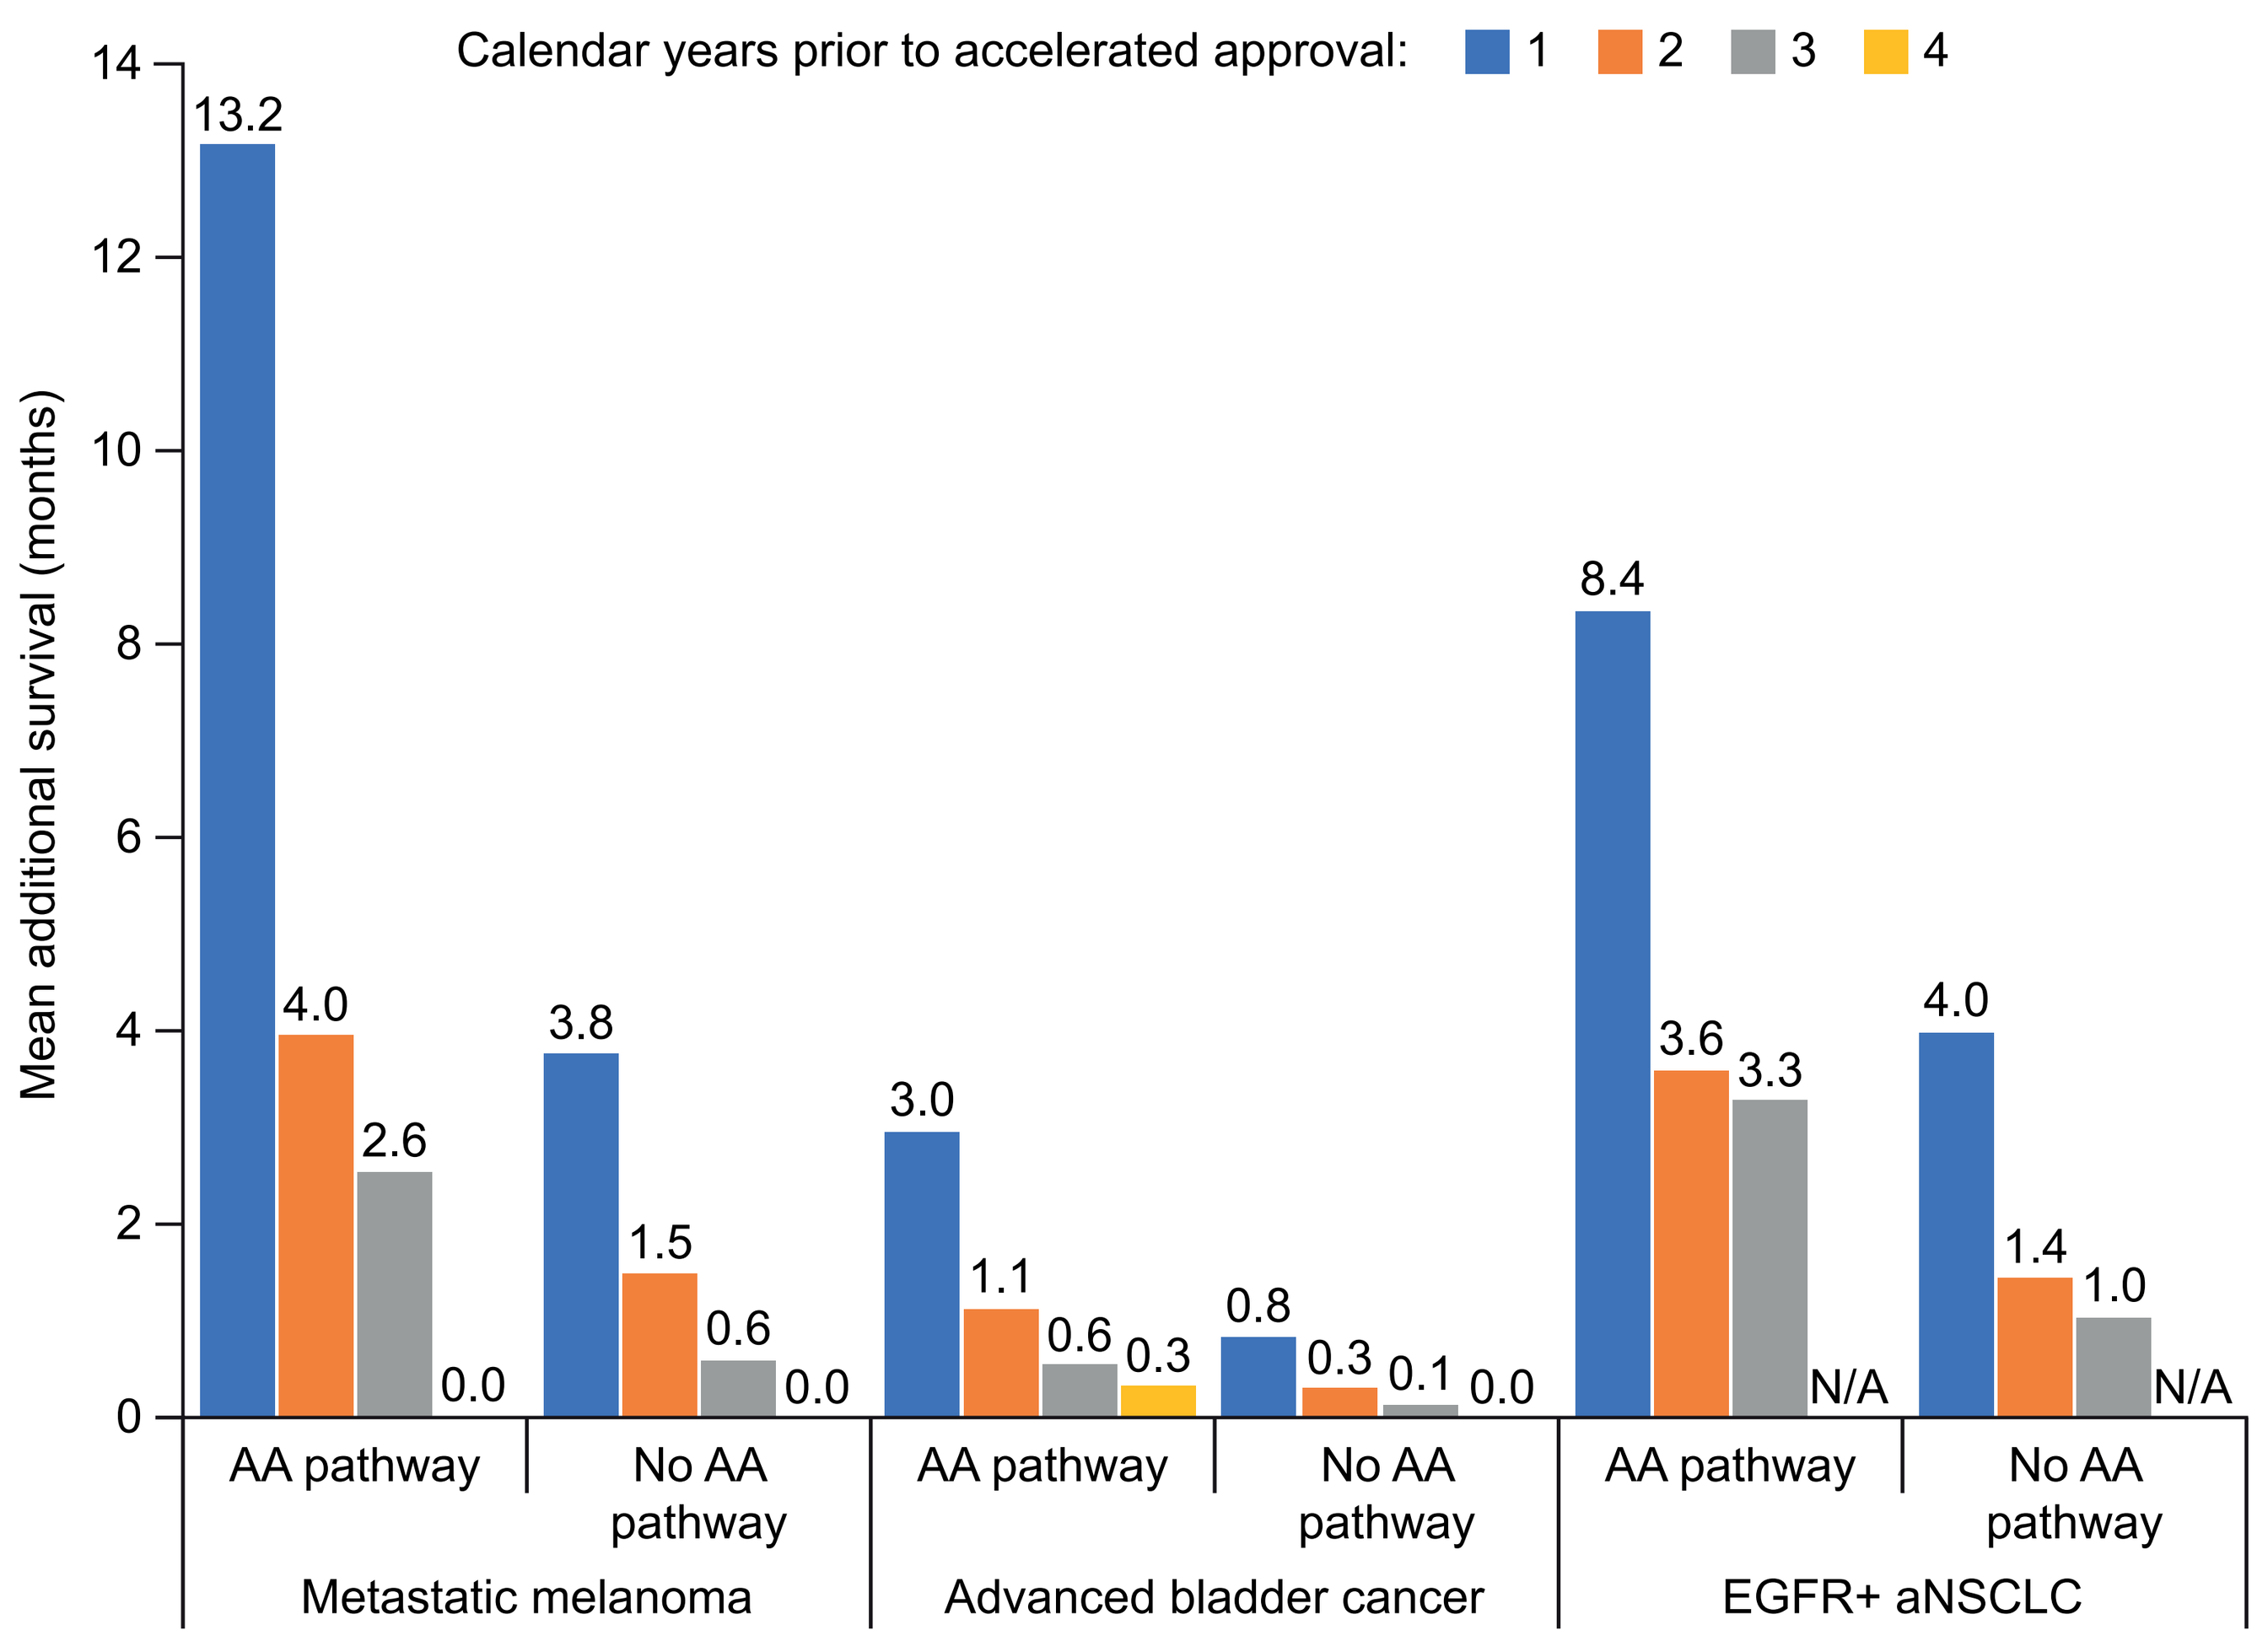
Calendar years prior to accelerated approval: 1 2 3 4
Mean additional survival (months)
14
12
10
8
6
4
2
0
13.2
4.0
2.6
0.0
3.8
1.5
0.6
0.0
3.0
1.1
0.6
0.3
0.8
0.3
0.1
0.0
8.4
3.6
3.3
N/A
4.0
1.4
1.0
N/A
AA pathway
No AA pathway
Metastatic melanoma
Advanced bladder cancer
EGFR+ aNSCLC

The calendar years prior to accelerated approval reflects the year of initial drug start relative to the accelerated approval date of the AA drug.
AA, accelerated approval; aNSCLC, advanced non-small cell lung cancer; EGFR+, epidermal growth factor receptor positive; N/A, not available.

CONCLUSIONS

- The AA drugs added additional survival to that of the initial drugs, with a majority of this additional survival being attributable to the AA pathway.
- The results were driven by both the effectiveness of the AA drug and the severity of the tumor and suggest that patients with tumors with rapid disease progression may stand to benefit more from shortening the time between innovations via the AA pathway.

DISCLOSURES

W. Wong, **T. To**, **A. Patel** and **S. Kowal** are employees of Genentech, Inc. and have stock in F. Hoffmann-La Roche AG. **D. Veenstra**, **M. Li** and **L. Garrison** received research funding for VeriTech Corporation from Genentech, Inc.

Funding statement: This study was sponsored by Genentech, Inc.
Acknowledgments: Medical writing assistance was provided by Rebecca Hornby, PhD, of Oxford PharmaGenesis, Oxford, UK, with funding from Genentech, Inc.

REFERENCES

1. FDA. Accelerated Approval Program. Available from: https://www.fda.gov/drugs/nda-and-bla-approvals/accelerated-approval-program (Accessed January 13, 2023).
2. Ladd V. The FDA's accelerated approval process is working. 2021. Available from: https://morningconsult.com/opinions/fdas-accelerated-approval-process-working/ (Accessed January 13, 2023).

Presented at ISPOR 2023, Boston, MA, USA, May 7–10, 2023