Abstract ID: 116006

Non-interventional, Real-world Study of Patients with Early Stage, Human Epidermal Growth Factor Receptor 2 (HER2) Positive Breast Cancer Receiving Biosimilar Trastuzumab

Shelbaya A[1], Kale H[2], Poorman GW[2], Inguva S[2], Alvir JM[3], Bair A[1], Stephens JM[2]

1. Pfizer Inc, Oncology Biosimilars HEOR, New York, NY, USA
2. OPEN Health, Bethesda, MD, USA
3. Pfizer Inc, Statistical Research and Data Science Center, New York, NY, USA

Objective

The objectives of this study were to understand the utilization patterns and safety profile of trastuzumab-qyyp and examine the healthcare resource use in patients prescribed trastuzumab-qyyp in a real-world setting.

Conclusions

- The current study showed that treatment utilization patterns and the safety profile of trastuzumab-qyyp among patients who initiated neoadjuvant or adjuvant treatment were consistent with the product label and the clinical recommendations.
- Healthcare resource use was substantial in this patient population, consistent with the substantial economic burden associated with breast cancer.

Presenting author: Dr. Ahmed Shelbaya

Email Dr. Shelbaya at Ahmed.Shelbaya@pfizer.com for more information

Click or scan this quick response (QR) to download this poster along with associated material.

References:

1. Smith J. U.S. Fda Approves Pfizer's Oncology Biosimilar Trazimera™ (Trastuzumab-Qyyp), A Biosimilar To Herceptin®1. In: Pfizer I, ed.: Pfizer, Inc; 2019.
2. Biegi T. Pfizer Receives European Approval For Oncology Biosimilar, Trazimera™ (Trastuzumab). In: Pfizer I, ed.: Pfizer, Inc; 2018.
3. Lammers PE, Dank M, Masetti R, et al. 154PDA randomized, double-blind study of PF-05280014 (a potential biosimilar) vs trastuzumab, both given with docetaxel (D) and carboplatin (C), as neoadjuvant treatment for operable human epidermal growth factor receptor 2-positive (HER2+) breast cancer. Annals of Oncology 2017;28(suppl_5):mdx362.005-mdx362.005.
4. Pegram M, Tan-Chiu E, Freyman A, et al. A randomized, double-blind study of PF-05280014 (a potential trastuzumab biosimilar) vs trastuzumab, both in combination with paclitaxel, as first-line treatment for HER2-positive metastatic breast cancer. ESMO2017.
5. Yin D, Barker KB, Li R, et al. A randomized phase 1 pharmacokinetic trial comparing the potential biosimilar PF-05280014 with trastuzumab in healthy volunteers (REFLECTIONS B327-01). British Journal of Clinical Pharmacology 2014;78(6):1281-90.

Acknowledgments

This study was sponsored by Pfizer. Copyright © 2022

Presented at the International Society for Pharmacoeconomics and Outcomes Research Annual Meeting • May 15–18, 2022 • Washington, DC, USA and Virtual.

Background

- Trazimera® (trastuzumab-qyyp) is a biosimilar to Herceptin® (trastuzumab) and is currently approved by the United States Food and Drug Administration[1] and the European Medicines Agency.[2]
- It is the fourth biosimilar to Herceptin® in Europe.
- In a randomized, non-inferiority, double-blind study, trastuzumab-qyyp was shown to be comparable to its originator in terms of the primary endpoint of pathological complete response as well as grade 3-4 adverse events.[3]
- However, there are no studies evaluating the treatment patterns and outcomes associated with trastuzumab-qyyp in a real-world setting.
- The objectives of this study were to understand the utilization of trastuzumab-qyyp and its benefits and risks in a real-world setting.

Methods

Study Design

An observational, multi-site study conducted in adult patients who received trastuzumab-qyyp as neoadjuvant or adjuvant therapy for the treatment of BC in Norway and the Netherlands was conducted.

Inclusion Criteria

- Patients with stage 0-3 HER2 positive breast cancer at the time of diagnosis or trastuzumab-qyyp initiation
- Initiated treatment with trastuzumab-qyyp for the neoadjuvant or adjuvant treatment of their disease and/or received at least 3 months of trastuzumab-qyyp treatment

Exclusion Criteria

- Age <18 years
- Stage 4 HER2 positive breast cancer at the time of diagnosis or trastuzumab-qyyp initiation
- active malignant disease requiring treatment with trastuzumab or currently on trastuzumab-emtasine

Data Source

- The patient's medical record served as the primary data source.
- Data was collected from 1 site in Norway and 6 sites in the Netherlands from February 2020 to July 2021.

Statistical Analysis

- Trastuzumab-qyyp treatment patterns, patient demographics, clinical characteristics, tolerability and healthcare resource utilization (HCRU) were assessed and presented descriptively.
- Healthcare resource utilization was assessed from 6 months prior to biosimilar initiation through to the informed consent date or the end of data collection period.

Results

Patient Demographics and Clinical Characteristics

- A total of 102 patients were included; 74 (72.6%) received trastuzumab-qyyp, 26 (25.5%) switched from Herceptin to trastuzumab-qyyp and 3 switched from other trastuzumab biosimilars.
- Demographic and clinical characteristics are presented in Table 1.

Table 1. Patient Demographics and Clinical Characteristics

Variable	Statistic or Category	All Patients (N=102)
Age (years)	Mean (SD)	56.3 (11.9)
Body Mass Index	Mean (SD)	26.8 (5.6)
Charlson comorbidity index	Mean (SD)	0.32 (0.96)
Healthcare coverage, N (%)	Public/Government	49 (48.0%)
	Private	31 (30.4%)
	Unknown	22 (21.6%)
Smoking status, N (%)	Current smoker	7 (6.9%)
	Former smoker	20 (19.6%)
	Never smoked	59 (57.8%)
	Unknown	16 (15.7%)
Family history of breast cancer, N (%)	Yes	36 (35.3%)
Menopausal status, N (%)	Premenopausal	27 (26.5%)
	Postmenopausal (due to LH-RH analog or oophorectomy)	3 (2.9%)
	Postmenopausal (not due to LH-RH analog or oophorectomy)	55 (53.9%)
	Unknown	17 (16.7%)
ER/PgR status, N (%)	ER + (>50% positive) / PgR +	51 (50.0%)
	ER + (<50% positive) / PgR +	1 (1.0%)
	ER + (>50% positive) / PgR -	16 (15.7%)
	ER + (<50% positive) / PgR -	4 (3.9%)
	ER – / PgR +	1 (1.0%)
	ER – / PgR -	29 (28.4%)
HER2 status, N (%)	HER2 positive (no further info. given)	39 (38.2%)
	HER2 positive: 2+ and CISH/FISH positive	16 (15.7%)
	HER2 positive: 3+	47 (46.1%)

Utilization of trastuzumab-qyyp

- The median trastuzumab-qyyp initial dose was 8.0 mg/kg, with dose change occurring in 78 (76.5%) patients. (Table 2)
- Trastuzumab-qyyp was part of combination therapy in 100 (98%) patients, where 38 (37.2%) combination regimens included platinum-based drugs and 40 (39.2%) regimens included pertuzumab. (Table 2)
- Further details on trastuzumab-qyyp utilization can be found in Table 2.

Table 2. Utilization and Patterns of trastuzumab-qyyp

Variable	Statistic or Category	All Patients (N=102)
Initial dose (mg/kg)	Mean (SD)	6.4 (1.9)
	Median (Q1-Q3)	8.0 (6.0–8.0)
Initial duration (minutes)	Mean (SD)	65.1 (47.6)
	Median (Q1-Q3)	35.0 (30.0 to 90.0)
Treatment duration (days)	Mean (SD)	245.6 (107.2)
Combination therapy, N (%)	Yes	100 (98.0%)
Type of combination therapy, N (%)	Chemotherapy	76 (74.5%)
	Hormone therapy	35 (34.3%)
	Immunotherapy	3 (2.9%)
	Radiation therapy	2 (2.0%)
	Chemotherapy + Immunotherapy	15 (14.7%)
	Chemotherapy + Radiation	1 (1.0%)
	Hormone therapy + Radiation	8 (7.8%)
Dose change, N (%)	Yes	78 (76.5%)

Figure 1. Healthcare Resource Utilization Among Patients

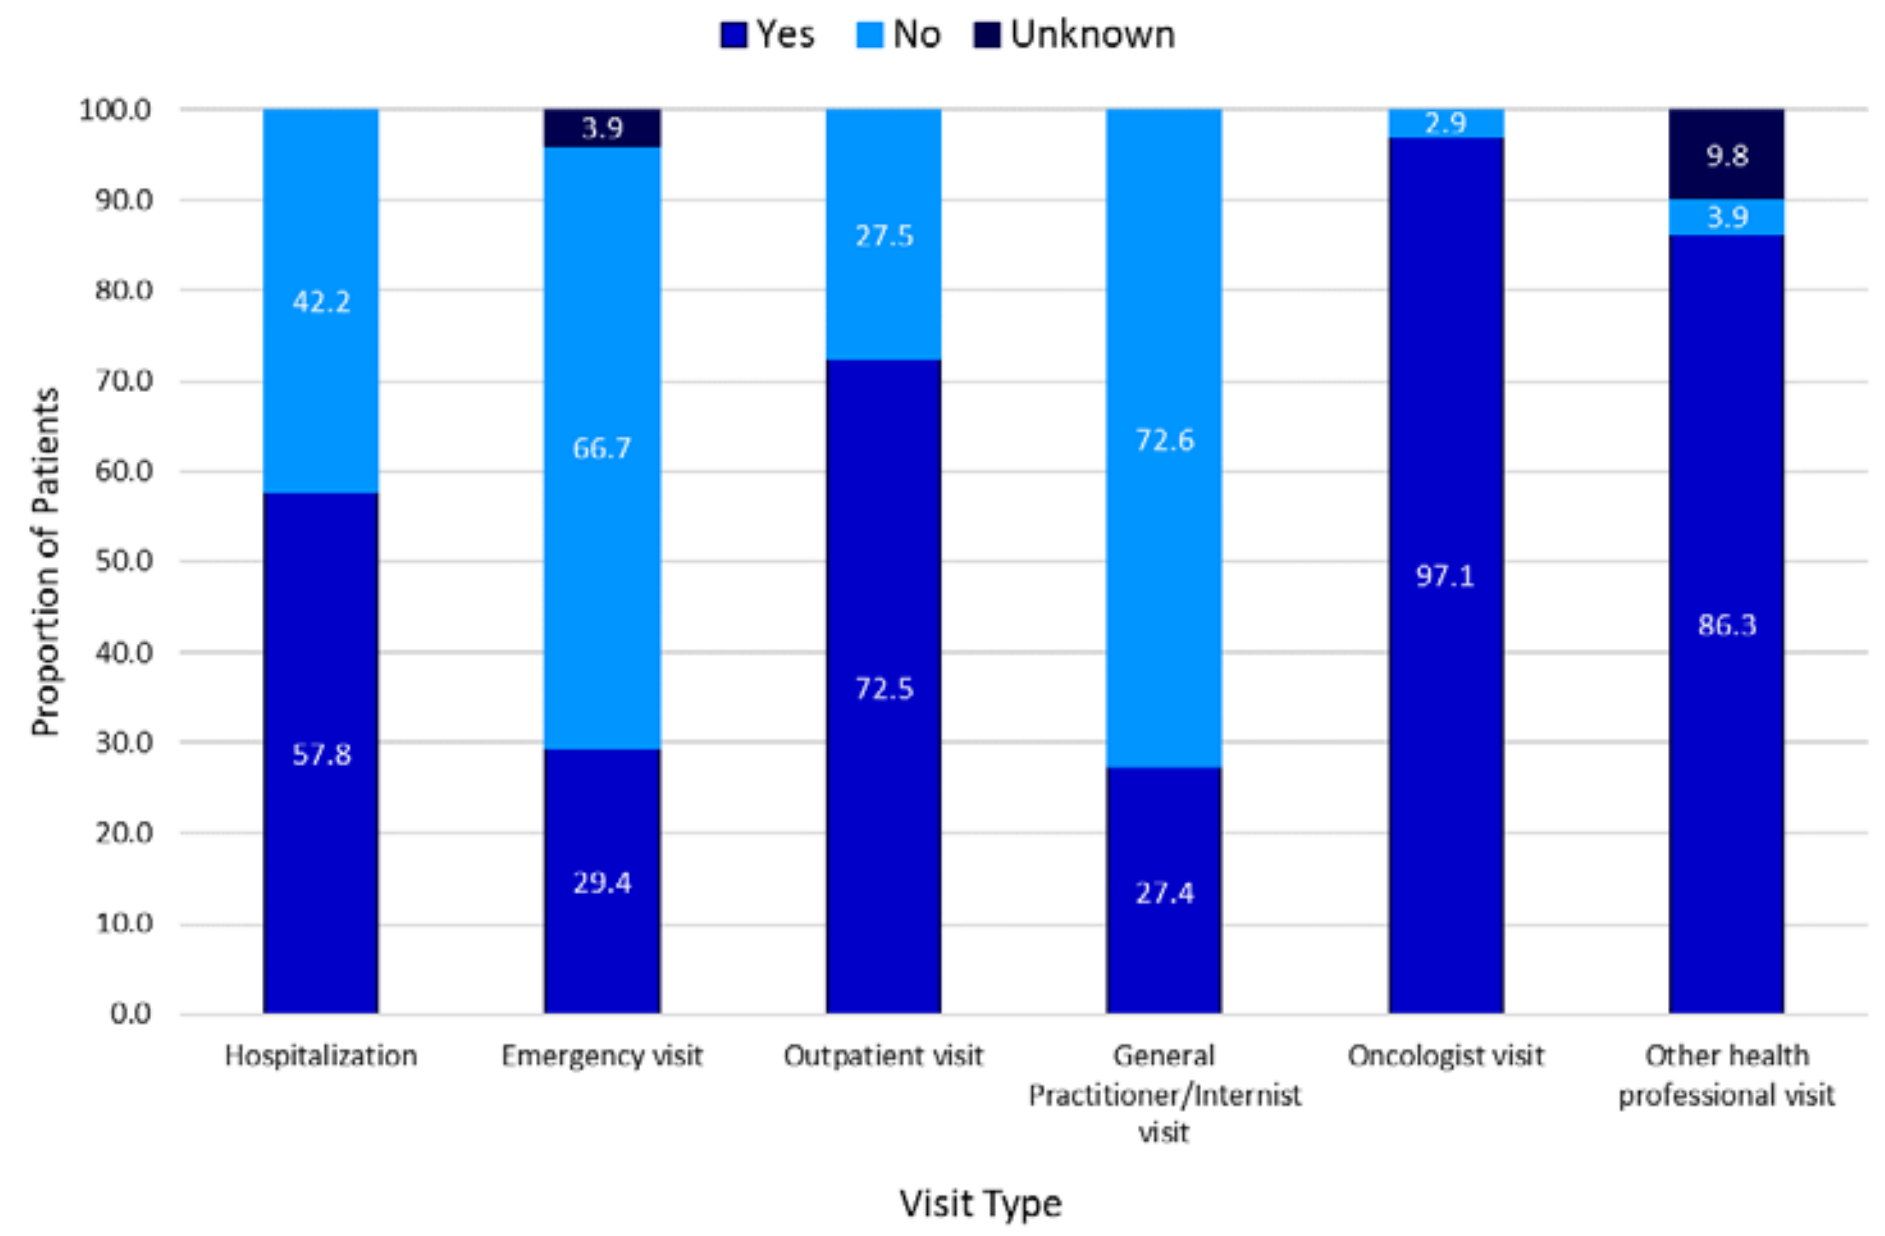
Yes
No
Unknown
100.0
90.0
80.0
70.0
60.0
50.0
40.0
30.0
20.0
10.0
0.0
Proportion of Patients
42.2
57.8
3.9
66.7
29.4
27.5
72.5
72.6
27.4
2.9
97.1
9.8
3.9
86.3
Hospitalization
Emergency visit
Outpatient visit
General Practitioner/Internist visit
Oncologist visit
Other health professional visit
Visit Type

Healthcare Resource Utilization

- Over 50% of the patients were hospitalized for a mean of 7.7 days.
- Reasons for hospitalization were breast cancer surgery including reconstructions (32.7%), PICC line/port-a-cath procedures (15.8%), fever (13.9%), ablation (5.9%), and other unspecified surgery (5.9%).
- Further details on the HCRU are presented in Figure 1.

Adverse Events and Cardiac Monitoring

- A total of 66 adverse events occurred and 38 (57.6%) were mild.
- Results associated with adverse events are presented in Table 3.
- Heart monitoring was conducted in 95 (93.1%) patients; 237 heart monitoring events recorded.
- Of 237 heart monitoring events, 221 (93.2%) had normal findings; trastuzumab-qyyp was stopped temporarily during 7 events and permanently during 1 event.

Table 3. Adverse Events During trastuzumab-qyyp Treatment (Event Level)

Variable	Statistic or Category	All Patients (N=102)
Severity/Grade, N (%)	Mild	38 (57.6%)
	Moderate	24 (36.4%)
	Severe	4 (6.1%)
trastuzumab-qyyp-Related, N (%)	Definitely	6 (9.1%)
	Probably	13 (19.7%)
	Unlikely	11 (16.7%)
	Unrelated	36 (54.6%)
Serious, N (%)	Yes	21 (31.8%)
Action Taken for trastuzumab-qyyp, N (%)	Withdrawn	16 (24.2%)
	Dose not changed	43 (65.1%)
	Not applicable	7 (10.6%)
Adverse Event Treatment, N (%)	None	30 (45.4%)
	Treatment with medications	32 (48.5%)
	Treatment without medication	4 (6.1%)
Outcome, N (%)	Resolved/recovered	58 (87.9%)
	Resolved/recovered with sequelae	4 (6.1%)
	Not resolved/not recovered	4 (6.1%)
	Fatal	0 (0.0%)

Copyright ©2021. All rights reserved.