Modeled Survival of People With Cystic Fibrosis Aged ≥6 Years Heterozygous for the *F508del* Mutation With a Minimal Function Mutation in Brazil Treated With Elexacaftor/Tezacaftor/Ivacaftor and Ivacaftor

Gabriel Ghizzi Pedra[1], Conor Daly[1], Leonardo Araujo Pinto[2], Andrea Lopez[3], Gabriela Vega-Hernandez[1], Jaime L. Rubin[3]

[1]Vertex Pharmaceuticals (Europe) Limited, London, UK; [2]Pontifícia Universidade Católica do Rio Grande do Sul, Porto Alegre, Rio Grande do Sul, Brazil; [3]Vertex Pharmaceuticals Incorporated, Boston, MA, USA

INTRODUCTION

- Cystic fibrosis (CF) is a rare genetic disease caused by mutations in the CF transmembrane conductance regulator (*CFTR*) gene. The progressive nature of the disease damages numerous organs, causing frequent complications, hospitalizations, and premature mortality[1-4]
- There is no cure for CF; however, CFTR modulators—small molecules that target the underlying cause of the disease—represent a major advancement in the treatment of CF[5-7]
- Available CFTR modulator therapies include ivacaftor (IVA), lumacaftor/ivacaftor (LUM/IVA), and tezacaftor/ivacaftor in combination with ivacaftor (TEZ/IVA), each of which is indicated to treat specific gene mutations
- In March 2022, regulatory approval of TRIKAFTA® (elexacaftor/tezacaftor/ivacaftor in combination with ivacaftor [ELX/TEZ/IVA]) was issued in Brazil to treat people with CF (pwCF) aged ≥6 years who have ≥1 *F508del* mutation in the *CFTR* gene[8]
- ELX/TEZ/IVA is a breakthrough therapy that has demonstrated robust and clinically meaningful benefits in pwCF, including those who are heterozygous for the *F508del* mutation and a minimal function mutation (*F*/MF), in whom previous CFTR modulator regimens were ineffective[9-11]
 - Phase 3/3b clinical trials have demonstrated unprecedented clinical benefits of ELX/TEZ/IVA, including improved lung function, respiratory symptoms, CFTR function, and nutritional outcomes[9-11]
 - A recent open-label extension study concluded that the rapid and robust improvements in lung function achieved with ELX/TEZ/IVA were maintained for up to 2 years of continued use in pwCF who initiated ELX/TEZ/IVA treatment at ages ≥12 years[12]
- In the absence of real-world studies that evaluate the survival and long-term health benefits of ELX/TEZ/IVA, simulation models have been developed to predict these outcomes in pwCF treated with ELX/TEZ/IVA in the US and UK.[13-16] However, the potential long-term benefits of ELX/TEZ/IVA in Brazil have not yet been evaluated

OBJECTIVE

- **To estimate the survival and long-term clinical benefits of adding ELX/TEZ/IVA to best supportive care (BSC) in pwCF aged ≥6 years with *F*/MF genotypes in Brazil, a population that did not have any CFTR modulator treatment options available before the approval of ELX/TEZ/IVA**

METHODS

Model Overview

- A person-level simulation model was developed to estimate the lifetime clinical benefits of ELX/TEZ/IVA + BSC compared with those of BSC alone in pwCF aged ≥6 years with *F*/MF genotypes in Brazil
 - Technical details of the model have been described previously[17]
 - The model structure was validated using real-world data from a long-term safety study in pwCF treated with ivacaftor over 5 years[18]; the 5-year survival projections from the model closely matched the 5-year mortality rates observed in ivacaftor-treated and -untreated pwCF from the study[19]
- A lifetime model horizon was used; a scenario analysis was conducted to evaluate clinical outcomes at 10 years
- Two simulated cohorts were assigned treatment with either ELX/TEZ/IVA + BSC or BSC alone; identical baseline characteristics were used for the 2 cohorts to ensure that any differences in outcomes were attributable to treatment (**Figure 1**)
- Projected survival estimates were based on background mortality hazards for Brazilian pwCF in the absence of CFTR modulator treatment derived from the Registro Brasileiro de Fibrose Cística (REBRAFC) 2016 annual report[20,21]
- Simulated individual survival estimates were calculated by combining the background survival mortality hazard with a Cox proportional hazards (CPH) model, using data from the US CF Foundation Patient Registry, which links survival to 9 risk factors[22]:
 - Age
 - Sex
 - Percent predicted forced expiratory volume in 1 second ($ppFEV_1$)
 - Weight-for-age *z*-score
 - *Staphylococcus aureus* respiratory infection
 - *Burkholderia cepacia* respiratory infection
 - Pancreatic sufficiency status
 - Diabetes
 - Annual number of pulmonary exacerbations (PEx)
- Individualized predictions of death are made during each cycle, conditional on the clinical characteristics identified by the CPH model; the probability of death was updated over time as the individual passed through the model and their characteristics were updated
- Differences in survival between pwCF receiving ELX/TEZ/IVA + BSC vs those receiving BSC alone are primarily driven by $ppFEV_1$, weight-for-age *z*-score and number of PEx, as ELX/TEZ/IVA is assumed to impact these 3 characteristics based on clinical evidence[9,10,12,21]

Figure 1. Schematic for ELX/TEZ/IVA Person-Level Simulation Model

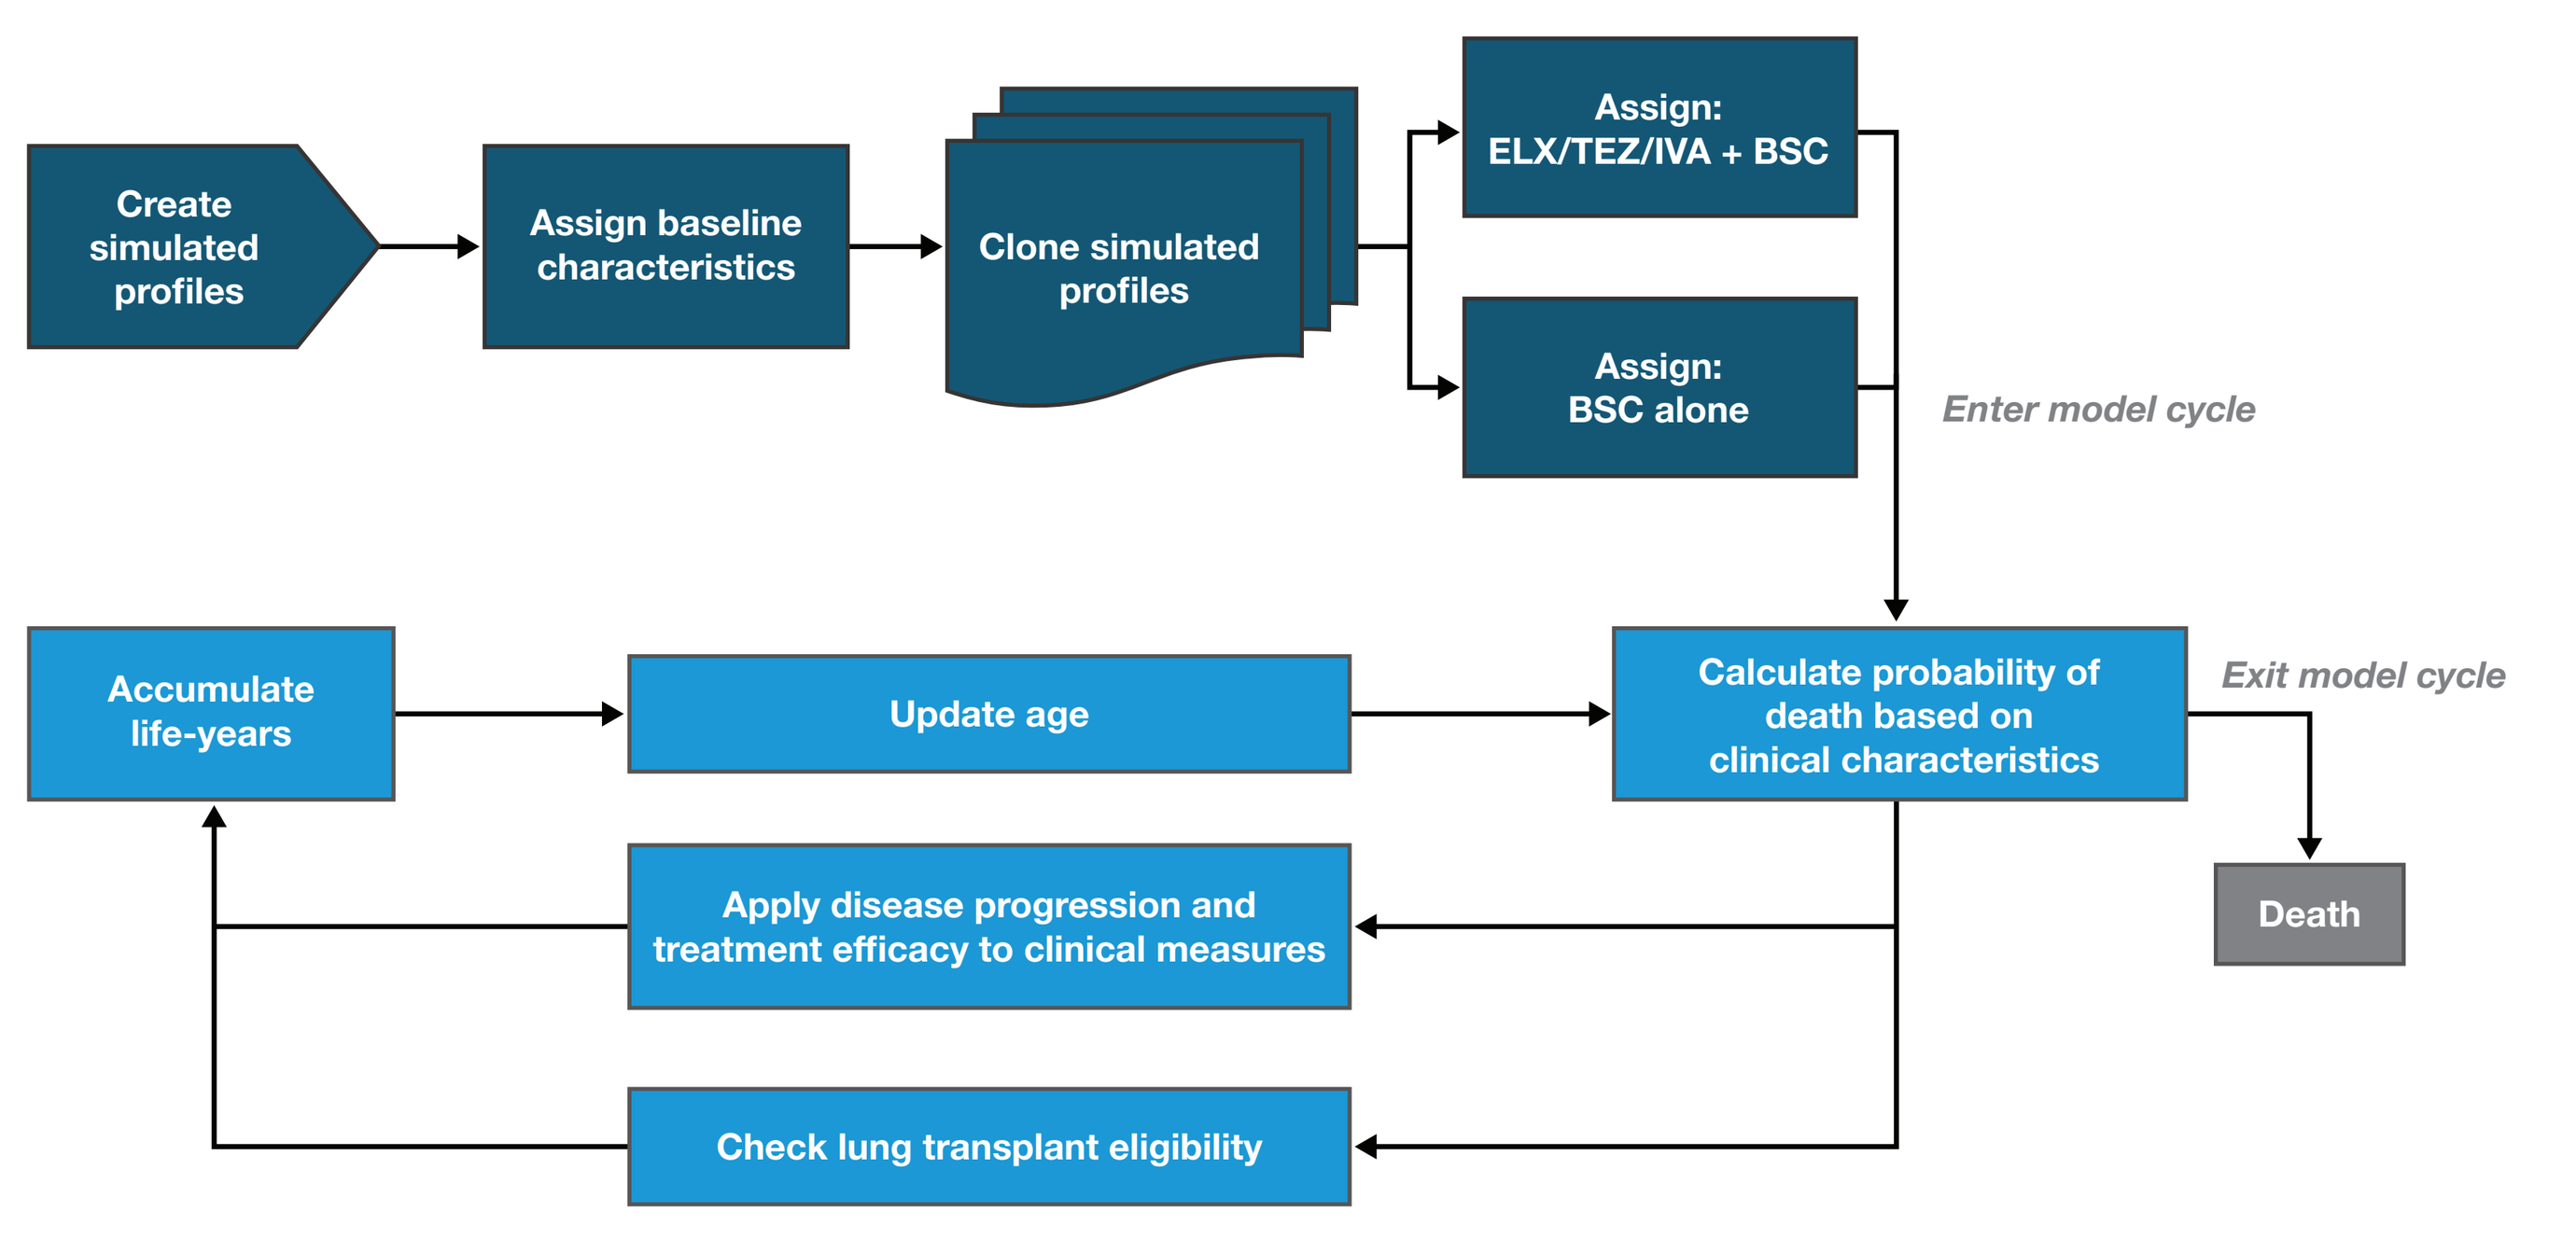
Create simulated profiles
Assign baseline characteristics
Clone simulated profiles
Assign: ELX/TEZ/IVA + BSC
Assign: BSC alone
Enter model cycle
Accumulate life-years
Update age
Calculate probability of death based on clinical characteristics
Exit model cycle
Death
Apply disease progression and treatment efficacy to clinical measures
Check lung transplant eligibility

BSC: best supportive care; ELX/TEZ/IVA: elexacaftor/tezacaftor/ivacaftor and ivacaftor.

Data Sources and Model Inputs

- Profiles for 2,000 simulated individuals aged ≥6 years were derived from baseline data from the Phase 3/3b clinical trials of ELX/TEZ/IVA that included pwCF with *F*/MF genotypes (Middleton 2019 for pwCF aged ≥12 years; Mall 2021 for pwCF aged 6-11 years),[9,10] weighted to match the age distribution of pwCF in the Brazilian CF registry (REBRAFC) 2018 annual report (age 6-11 years, 35.8%; age ≥12 years, 64.2%)[23]
 - To examine the impact of early ELX/TEZ/IVA treatment initiation, the subset of profiles for pwCF aged 6-11 years was simulated
- Background disease progression inputs were derived from published literature, and ELX/TEZ/IVA clinical efficacy inputs were informed by clinical trial data (**Table 1**)

Table 1. Model Inputs for Disease Progression and ELX/TEZ/IVA Treatment Efficacy

Parameter	Time Period	BSC Alone	ELX/TEZ/IVA Initiated During Ages 6-11 Years	ELX/TEZ/IVA Initiated During Ages ≥12 Years
Acute change in $ppFEV_1$ from baseline	Weeks 0-24	0.0	11.0 percentage point increase[9]	14.3 percentage point increase[10]
Annual change in $ppFEV_1$	Weeks 24+	Age-specific rates[24] Ages 6-8 years: –1.32 Ages 9-12 years: –1.32 Ages 13-17 years: –2.37 Ages 18-24 years: –2.52 Ages 25+ years: –1.86	90% reduction relative to BSC alone (conservative assumption based on maintenance of lung function improvements observed over 2 years in the open-label extension study)[12] Ages 6-8 years: –0.132 Ages 9-12 years: –0.132 Ages 13-17 years: –0.237 Ages 18-24 years: –0.252 Ages 25+ years: –0.186	
Rate of PEx requiring IV antibiotics and/or hospitalization	Lifetime	Predicted rate is conditional on age and $ppFEV_1$[25,26]	No impact assumed during ages 6-11 years; 78% lower rate relative to BSC alone applied after pwCF turn ≥12 years[10,12]	78% lower rate relative to BSC alone[10,12]
Mean change in weight-for-age *z*-score from baseline	Weeks 0-24	0.0	0.23-unit increase[9,21]	0.30-unit increase[10,21]

BSC: best supportive care; ELX/TEZ/IVA: elexacaftor/tezacaftor/ivacaftor and ivacaftor; IV: intravenous; PEx: pulmonary exacerbation; $ppFEV_1$: percent predicted forced expiratory volume in 1 second.

- It is assumed that 24.5% of simulated individuals receive a lung transplant when falling below the $ppFEV_1$ threshold of 30 percentage points,[27] based on transplant rates among pwCF in the UK[28]
- To evaluate the impact of ELX/TEZ/IVA on pwCF who remain on therapy and because there were few (<2%) discontinuations in the 24-week clinical trials and subsequent extension studies, all analyses assumed 100% persistence (ie, no ELX/TEZ/IVA discontinuations)

RESULTS

- In the simulated cohort, the mean baseline age was 20.1 years and the mean baseline $ppFEV_1$ was 71.8 percentage points
- Treatment with ELX/TEZ/IVA + BSC was projected to increase the median survival by 35.4 years compared with treatment with BSC alone in Brazilian pwCF aged ≥6 years with *F*/MF genotypes (67.6 vs 32.2 years) (**Figure 2**)

Figure 2. Projected Impact of ELX/TEZ/IVA + BSC on Survival in pwCF Aged ≥6 Years

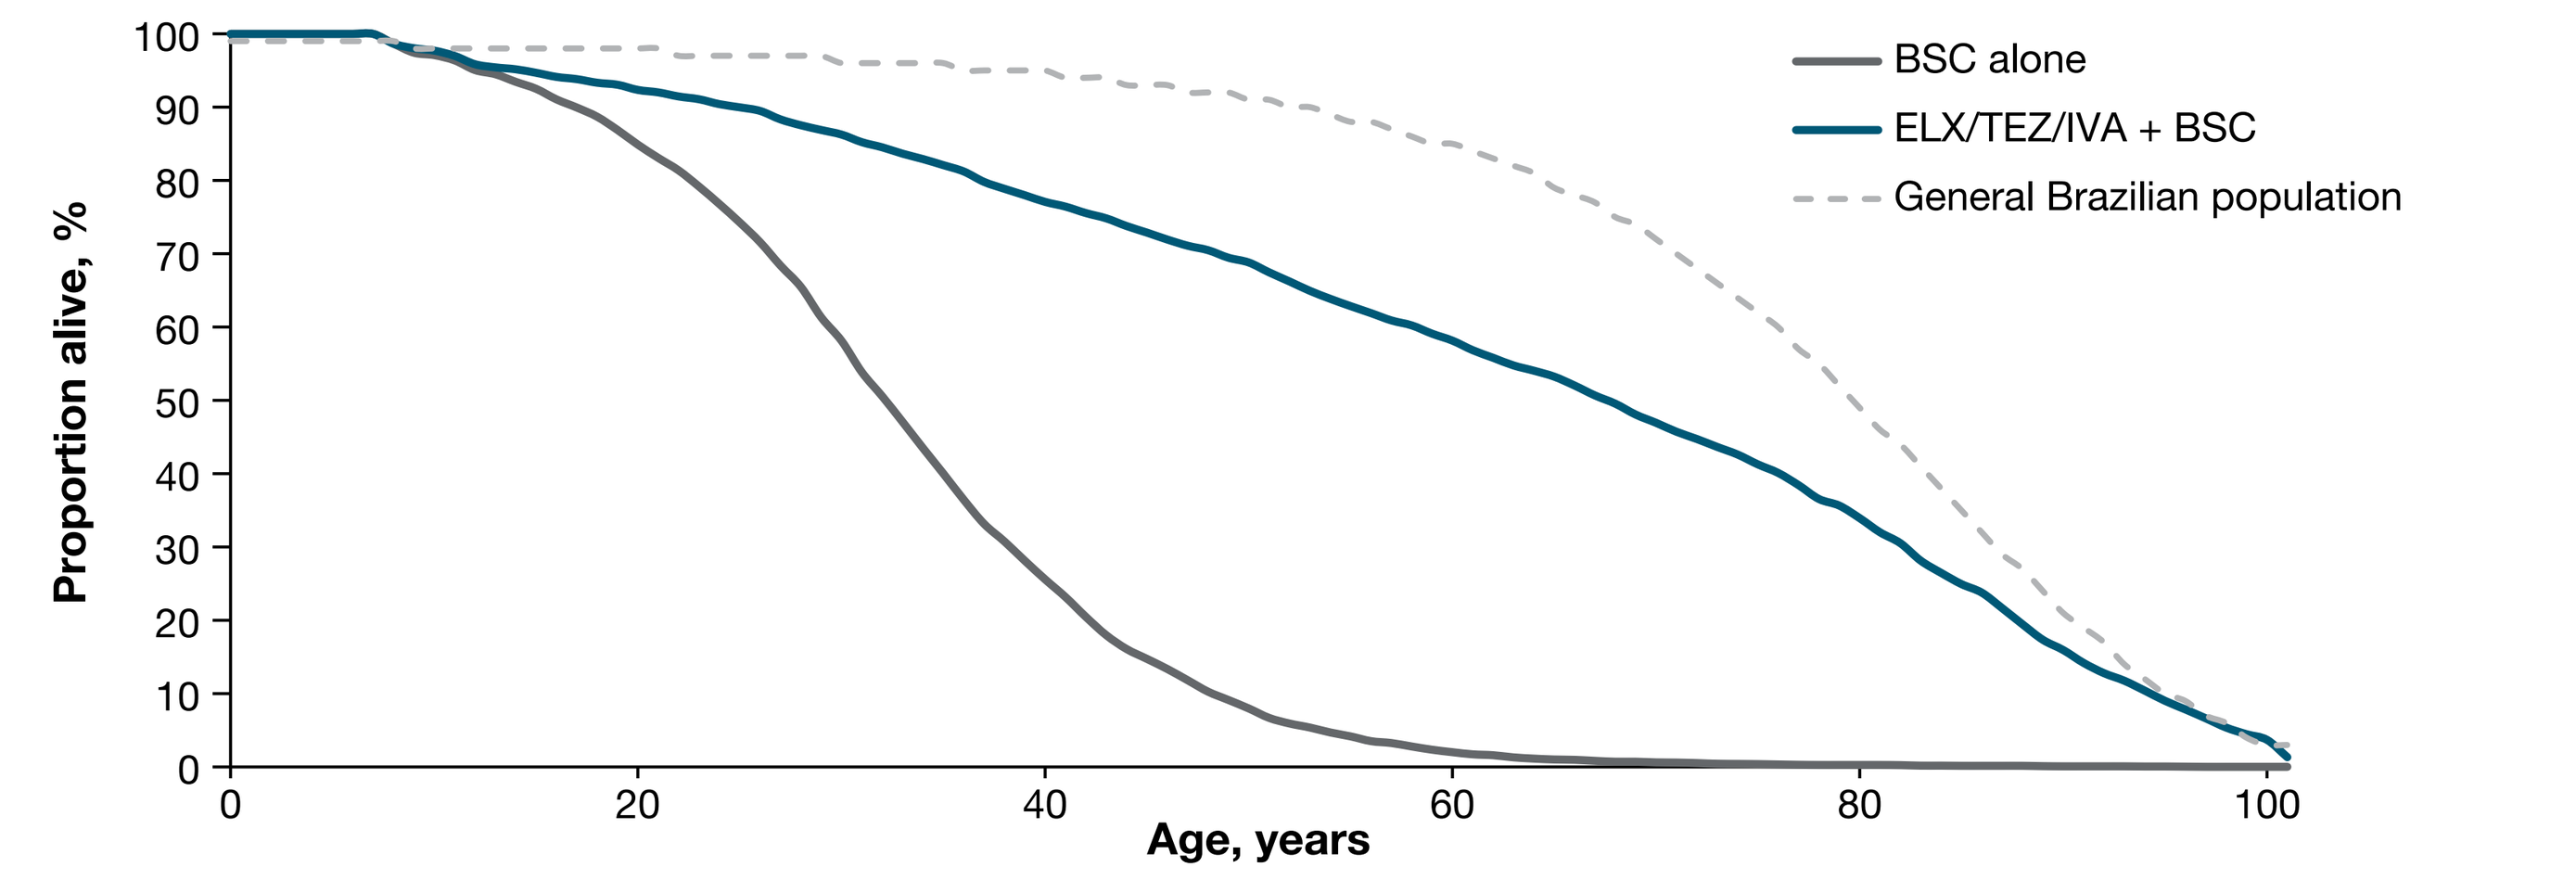
Proportion alive, %
100
90
80
70
60
50
40
30
20
10
0
0
20
40
60
80
100
Age, years
BSC alone
ELX/TEZ/IVA + BSC
General Brazilian population

BSC: best supportive care; ELX/TEZ/IVA: elexacaftor/tezacaftor/ivacaftor and ivacaftor; pwCF: people with CF.

- In addition to prolonging survival, treatment with ELX/TEZ/IVA + BSC was projected to provide substantial additional clinical benefit vs treatment with BSC alone (**Table 2**), including:
 - Increase in the proportion of life-years spent with mild disease ($ppFEV_1$ ≥70) nearly 3-fold
 - Reduction in the number of PEx requiring IV antibiotics or hospitalization over a lifetime by 66%
 - Elimination of the need for lung transplant among pwCF treated with ELX/TEZ/IVA + BSC
- Survival gains increased when initiating treatment at a younger age; those starting ELX/TEZ/IVA + BSC at ages 6-11 years were projected to live 42.1 years longer than those receiving BSC alone (**Figure 3**)

Table 2. Projected Impact of ELX/TEZ/IVA + BSC on Clinical Outcomes in pwCF Aged ≥6 Years Over Lifetime Horizon

	BSC	ELX/TEZ/IVA	Δ ELX/TEZ/IVA – BSC
Survival			
Median predicted survival, years	32.2	67.6	**+35.4**
Mean residual life-years (ie, years since model entry)	16.5	46.3	**+29.8**
Other clinical outcomes			
Mean change in $ppFEV_1$, %	–29	5	**+33**
Proportion of life-years spent with:			
$ppFEV_1$ >70, %	26	69	**+43**
$ppFEV_1$ 40-70, %	44	31	**–14**
$ppFEV_1$ <40, %	29	0	**–29**
Total lifetime number of PEx	16.4	5.6	**–10.9**
Annual rate of PEx (per person-year)	1.1	0.1	**–0.9**
Proportion undergoing lung transplant, %	7	0	**–7**

BSC: best supportive care; ELX/TEZ/IVA: elexacaftor/tezacaftor/ivacaftor and ivacaftor; PEx: pulmonary exacerbation; $ppFEV_1$: percent predicted forced expiratory volume in 1 second; pwCF: people with CF.

Figure 3. Impact of ELX/TEZ/IVA Age of Initiation on Projected Survival

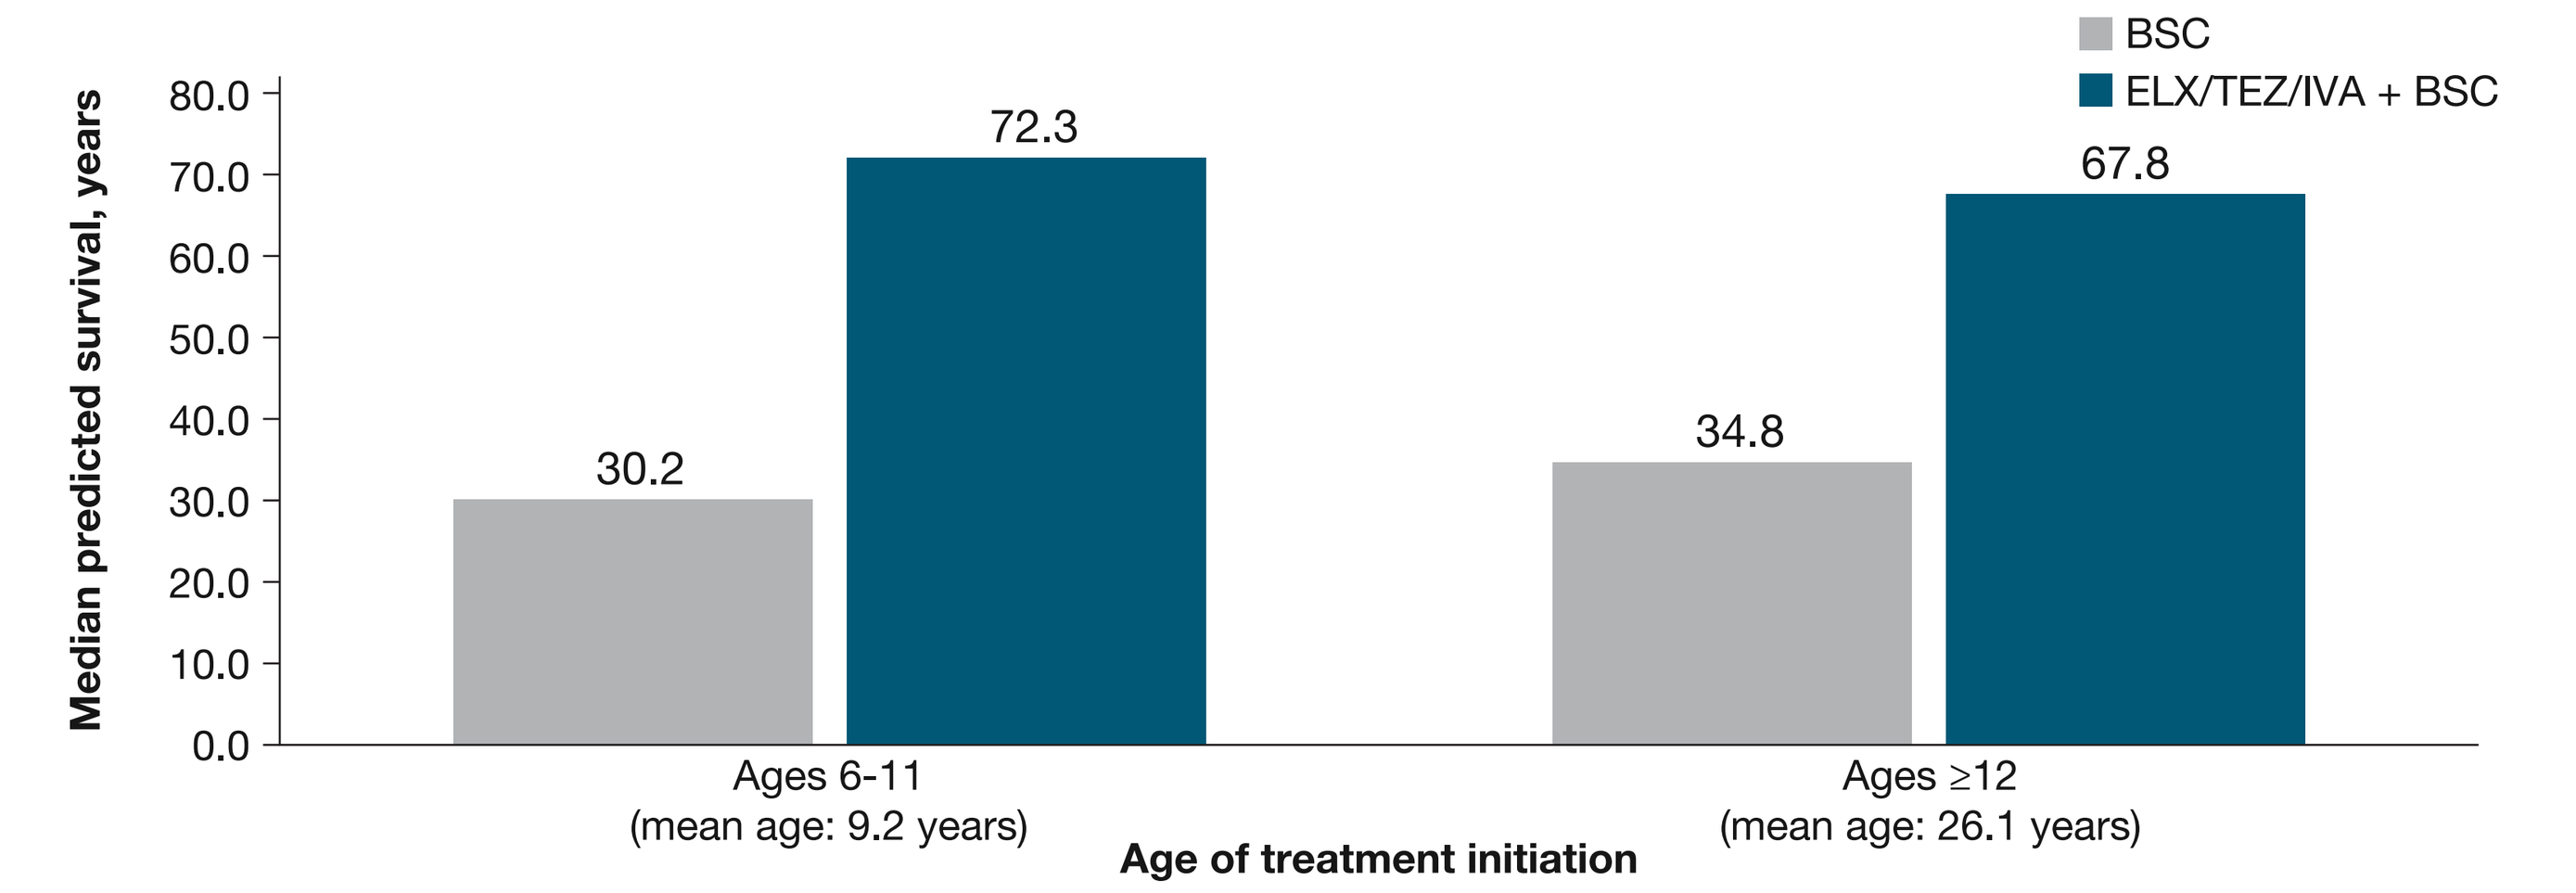
BSC
ELX/TEZ/IVA + BSC
Median predicted survival, years
80.0
70.0
60.0
50.0
40.0
30.0
20.0
10.0
0.0
30.2
72.3
34.8
67.8
Ages 6-11
(mean age: 9.2 years)
Ages ≥12
(mean age: 26.1 years)
Age of treatment initiation

BSC: best supportive care; ELX/TEZ/IVA: elexacaftor/tezacaftor/ivacaftor and ivacaftor.

- At 10 years, 18% more pwCF treated with ELX/TEZ/IVA + BSC were alive than those treated with BSC alone (89% vs 72%)
- After 10 years of treatment, the $ppFEV_1$ for pwCF treated with ELX/TEX/IVA is expected to be nearly 27 percentage points higher than the $ppFEV_1$ for those treated with BSC alone (**Table 3**)

Table 3. Projected Impact of ELX/TEZ/IVA + BSC on Clinical Outcomes in pwCF Aged ≥6 Years Over 10-Year Time Horizon

	BSC	ELX/TEZ/IVA	Δ ELX/TEZ/IVA – BSC
Survival			
Proportion alive at the end of the 10-year horizon, %	72	89	**+18**
Mean residual life-years (ie, years since model entry)	8.8	9.5	**+0.7**
Other clinical outcomes			
Mean change in $ppFEV_1$, %	–15	11	**+27**
Proportion of life-years spent with:			
$ppFEV_1$ >70, %	40	72	**+32**
$ppFEV_1$ 40-70, %	45	28	**–18**
$ppFEV_1$ <40, %	14	0	**–14**
Total lifetime number of PEx	7.4	1.3	**–6.1**
Annual rate of PEx (per person-year)	0.8	0.1	**–0.7**
Proportion undergoing lung transplant, %	3	0	**–3**

BSC: best supportive care; ELX/TEZ/IVA: elexacaftor/tezacaftor/ivacaftor and ivacaftor; PEx: pulmonary exacerbation; $ppFEV_1$: percent predicted forced expiratory volume in 1 second; pwCF: people with CF.

LIMITATIONS

- Where Brazilian genotype-specific data were not available, model inputs were based on studies conducted in other genotype populations and/or countries (eg, the annual rate of $ppFEV_1$ decline was based on a study conducted in pwCF homozygous for *F508del* in the US[24])
- Treatment effects of ELX/TEZ/IVA + BSC were based on published clinical data for up to 2 years of treatment;[9-12] assumptions beyond this period were extrapolated

CONCLUSIONS

- ELX/TEZ/IVA + BSC is projected to **increase survival by over 3 decades** in pwCF aged ≥6 years with *F*/MF genotypes in Brazil, a group that did not have a previous CFTR modulator treatment option
- Treatment with ELX/TEZ/IVA + BSC is also projected to provide substantial clinical benefits compared with BSC alone, including a greater proportion of lifetime with higher lung function, fewer PEx, and lower risk of lung transplant
- **Earlier initiation of treatment** may prevent long-term disease progression and mortality; model projections suggest that initiation during ages 6-11 years **may add approximately 4 decades of survival benefit** vs BSC alone, allowing these pwCF to achieve near-normal life expectancy
- The projected survival benefits of this magnitude together with the fact that pwCF treated with ELX/TEZ/IVA + BSC are projected to spend a greater proportion of their lifetime with mild disease ($ppFEV_1$ ≥70) are encouraging and suggest that ELX/TEZ/IVA + BSC treatment could have the potential to transform health outcomes for many people living with CF

REFERENCES

1. Castellani C, Assael BM. *Cell Mol Life Sci.* 2017;74(1):129-40.
2. CFF. Cystic Fibrosis Foundation (CFF). https://www.cff.org/What-is-CF/About-Cystic-Fibrosis/. Accessed April 15, 2022.
3. Dechecchi MC, et al. *Ann Transl Med.* 2018;6(17):334.
4. O'Sullivan BP, Freedman SD. *Lancet.* 2009;373(9678):1891-904.
5. Clancy JP. *Pediatr Pulmonol.* 2018;53(S3):S4-S11.
6. Paranjape SM, Mogayzel PJ Jr. *Paediatr Respir Rev.* 2018;25:64-72.
7. Taylor-Cousar JL, et al. *ERJ Open Res.* 2019;5(2):00082-2019.
8. Agência Nacional de Vigilância Sanitária (Anvisa). https://www.gov.br/anvisa/pt-br/assuntos/medicamentos/novos-medicamentos-e-indicacoes/trikafta-r-elexacaftor-tezacaftor-ivacaftor-novo-registro. Accessed March 1, 2022.
9. Mall S, et al. Poster presented at the 24th German Cystic Fibrosis Conference (DMT); Virtual; November 19-20, 2021.
10. Middleton PG, et al. *N Engl J Med.* 2019;381(19):1809-19.
11. Zemanick ET, et al. *Am J Respir Crit Care Med.* 2021;203(12):1522-32.
12. Daines CL, et al. Poster presented at the 35th Annual North American Cystic Fibrosis CF Conference; Virtual; November 1-5, 2021.
13. Lopez A, et al. Poster presented at AMCP Annual Meeting; Virtual; April 21-24, 2020.
14. Lopez A, et al. Poster presented at AMCP Nexus 2021; Denver, CO; October 18-21, 2021.
15. Vega-Hernandez G, et al. Poster presented at the Professional Society for Health Economics and Outcomes Research (ISPOR); Virtual; May 17-20, 2021.
16. Vega-Hernandez G, et al. Poster presented at the Professional Society for Health Economics and Outcomes Research (ISPOR); Virtual; May 17-20, 2021.
17. Rubin JL, et al. *Ther Adv Respir Dis.* 2019;13:1-23.
18. Volkova N, et al. *J Cyst Fibros.* 2020;19(1):68-79.
19. McGarry L, et al. Oral presentation at the Professional Society for Health Economics and Outcomes Research (ISPOR); Virtual; May 16-20, 2020.
20. Registro Brasileiro de Fibrose Cística 2016. http://portalgbefc.org.br/ckfinder/userfiles/files/REBRAFC_2016.pdf. Accessed October 25, 2021.
21. Data on file. Vertex Pharmaceuticals Incorporated, Boston, MA.
22. Liou TG, et al. *Am J Epidemiol.* 2001;153(4):345-52.
23. Registro Brasileiro de Fibrose Cística 2018. http://portalgbefc.org.br/ckfinder/userfiles/files/REBRAFC_2018.pdf. Accessed October 25, 2021.
24. Sawicki GS, et al. Poster presented at the American Thoracic Society International Conference; Washington, DC; May 19-24, 2017.
25. Goss CH, Burns, JL. *Thorax.* 2007;62(4):360-7.
26. Whiting P, et al. *Health Technol Assess.* 2014;18(18):1-106.
27. Maurer JR, et al. *Transplantation.* 1998;66(7):951-6.
28. Cystic Fibrosis Trust. UK Cystic Fibrosis Registry Annual Data Report 2019. https://www.cysticfibrosis.org.uk/sites/default/files/2020-12/2019%20Registry%20Annual%20Data%20report_Sep%202020.pdf. Accessed April 15, 2022.

AUTHOR DISCLOSURES

GGP, CD, AL, GH-V and **JLR** are employees of Vertex Pharmaceuticals Incorporated and may own stocks or stock options in that company.

ACKNOWLEDGMENTS

Editorial coordination and support were provided by Nathan Blow, PhD, who is an employee of Vertex Pharmaceuticals Incorporated and may own stock or stock options in that company. Editorial support was provided by Jackie Highland, PhD, of ArticulateScience LLC under the guidance of the authors and funded by Vertex Pharmaceuticals.

Presented at the ISPOR 2022 Annual Conference, Washington, DC area and virtual, 15-18 May 2022.

Sponsored by Vertex Pharmaceuticals Incorporated