Certolizumab Pegol Persistence and Adherence by Drug Formulation, in Patients with Axial Spondyloarthritis, Psoriatic Arthritis, or Rheumatoid Arthritis

Silky Beaty,[1] Miriam L. Zichlin,[2] Sari Hopson,[1,*] Francesca Devine,[2] Selina Pi,[2] Josiah Moore,[3] Elyse Swallow[2]

Presented at ISPOR 2022 | May 15–18 | National Harbor, MD

Objectives

Primary: To characterize the demographic and clinical characteristics of patients who initiate certolizumab pegol (CZP), a biologic administered as a lyophilized powder (LYO) in-office or as a prefilled syringe (PFS) at home, by formulation, and to assess persistence and adherence to CZP by formulation.

Secondary: To evaluate the association between baseline biologic therapy adherence and CZP formulation initiation (i.e., LYO vs. PFS) among biologic-experienced patients.

Background

- Adherence to biologic therapy, such as CZP, a PEGylated Fc-free tumor necrosis factor inhibitor, remains a significant problem in the treatment of rheumatologic diseases.[1,2]
- CZP comes in the form of a LYO powder that is administered by a healthcare provider as an in-office injection or in the form of a PFS solution that is administered by the patient or a caregiver as an at-home injection.

Methods

- Adult patients with ≥1 prescription for CZP from January 1st, 2015 to March 31st, 2020 (first CZP prescription = index date), ≥1 diagnosis of axial spondyloarthritis (axSpA), psoriatic arthritis (PsA), or rheumatoid arthritis (RA) in the 12-months prior to the index date (baseline), and continuous eligibility during baseline were identified from the IBM® MarketScan® database.
- Baseline characteristics were evaluated and stratified by CZP formulation. In addition, demographics, clinical characteristics, and biologic adherence during baseline were compared between CZP formulations.
- Among patients in the sample with ≥1 biologic therapy during the baseline period and who initiated CZP LYO at index, univariate and multivariate generalized linear regressions were estimated.
- Persistence was calculated as the number of days from index date through the earliest date of CZP discontinuation (the beginning of a 90-day period without a CZP prescription), formulation switch, or end of eligibility.
- Adherence was calculated as the number of observed CZP claims divided by the number of expected CZP claims (determined based on package insert dosing instructions) during the persistent period and reported as a percentage.

Results

- 4,014 (LYO=1,437; PFS=2,577) patients who initiated CZP following a diagnosis for axSpA, PsA, or RA were identified **(Figure 1)**.
- Patient demographics and baseline characteristics in the two cohorts are reported in **Table 1**.
 - Among patients with axSpA, PsA, or RA, nearly two-thirds initiated the PFS formulation of CZP and resided in the South region.
 - Patients in the CZP LYO cohort were older and had more comorbidities.
- For both cohorts, median CZP persistence was ~9 months (LYO: 281 days; PFS: 286 days). More than three-quarters (LYO: 77.45%; PFS: 76.02%) of patients had adherence ≥80% **(Summary)**.
- Two-thirds of all patients received ≥1 biologic throughout the 12-month baseline period, though more patients initiating CZP PFS were biologic-experienced compared to those initiating CZP LYO. Across all indications, adalimumab and etanercept were the most common biologics received during baseline **(Figure 2)**.
- High adherence was seen to the biologic therapy received immediately prior to CZP during baseline; however, more patients initiating CZP LYO were adherent to the biologic prior to CZP initiation during baseline than patients initiating CZP PFS **(Figure 3)**.
- More patients who initiated CZP LYO received their most recent prior biologic therapy in-office during the baseline period, compared to patients who initiated CZP PFS **(Figure 4)**.
- Among biologic-experienced patients, there was no association between prior biologic adherence and initiation of CZP LYO after adjustment for demographic and clinical characteristics. However, increased age, female sex, fewer biologics received during the baseline period, and office administration of the biologic received immediately prior to CZP were significantly associated with the initiation of CZP LYO as compared to initiation of CZP PFS **(Table 2)**.
- Similar results were observed across indications.

Summary

Persistence and adherence to CZP was high, and similar by formulation, either

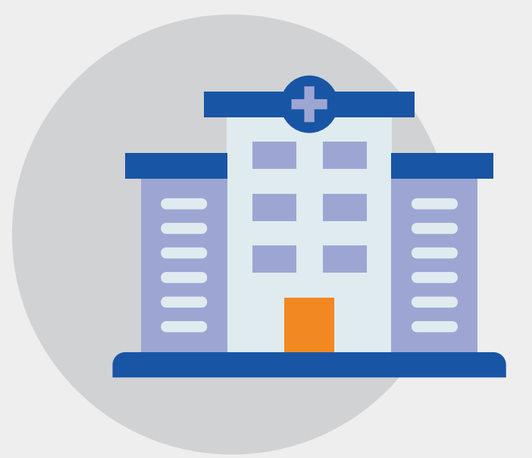

Lyophilized Powder (LYO)

or

Prefilled Syringe (PFS)

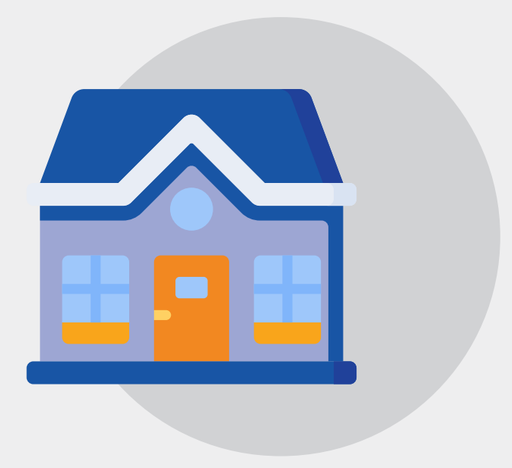

Persistence by formulation

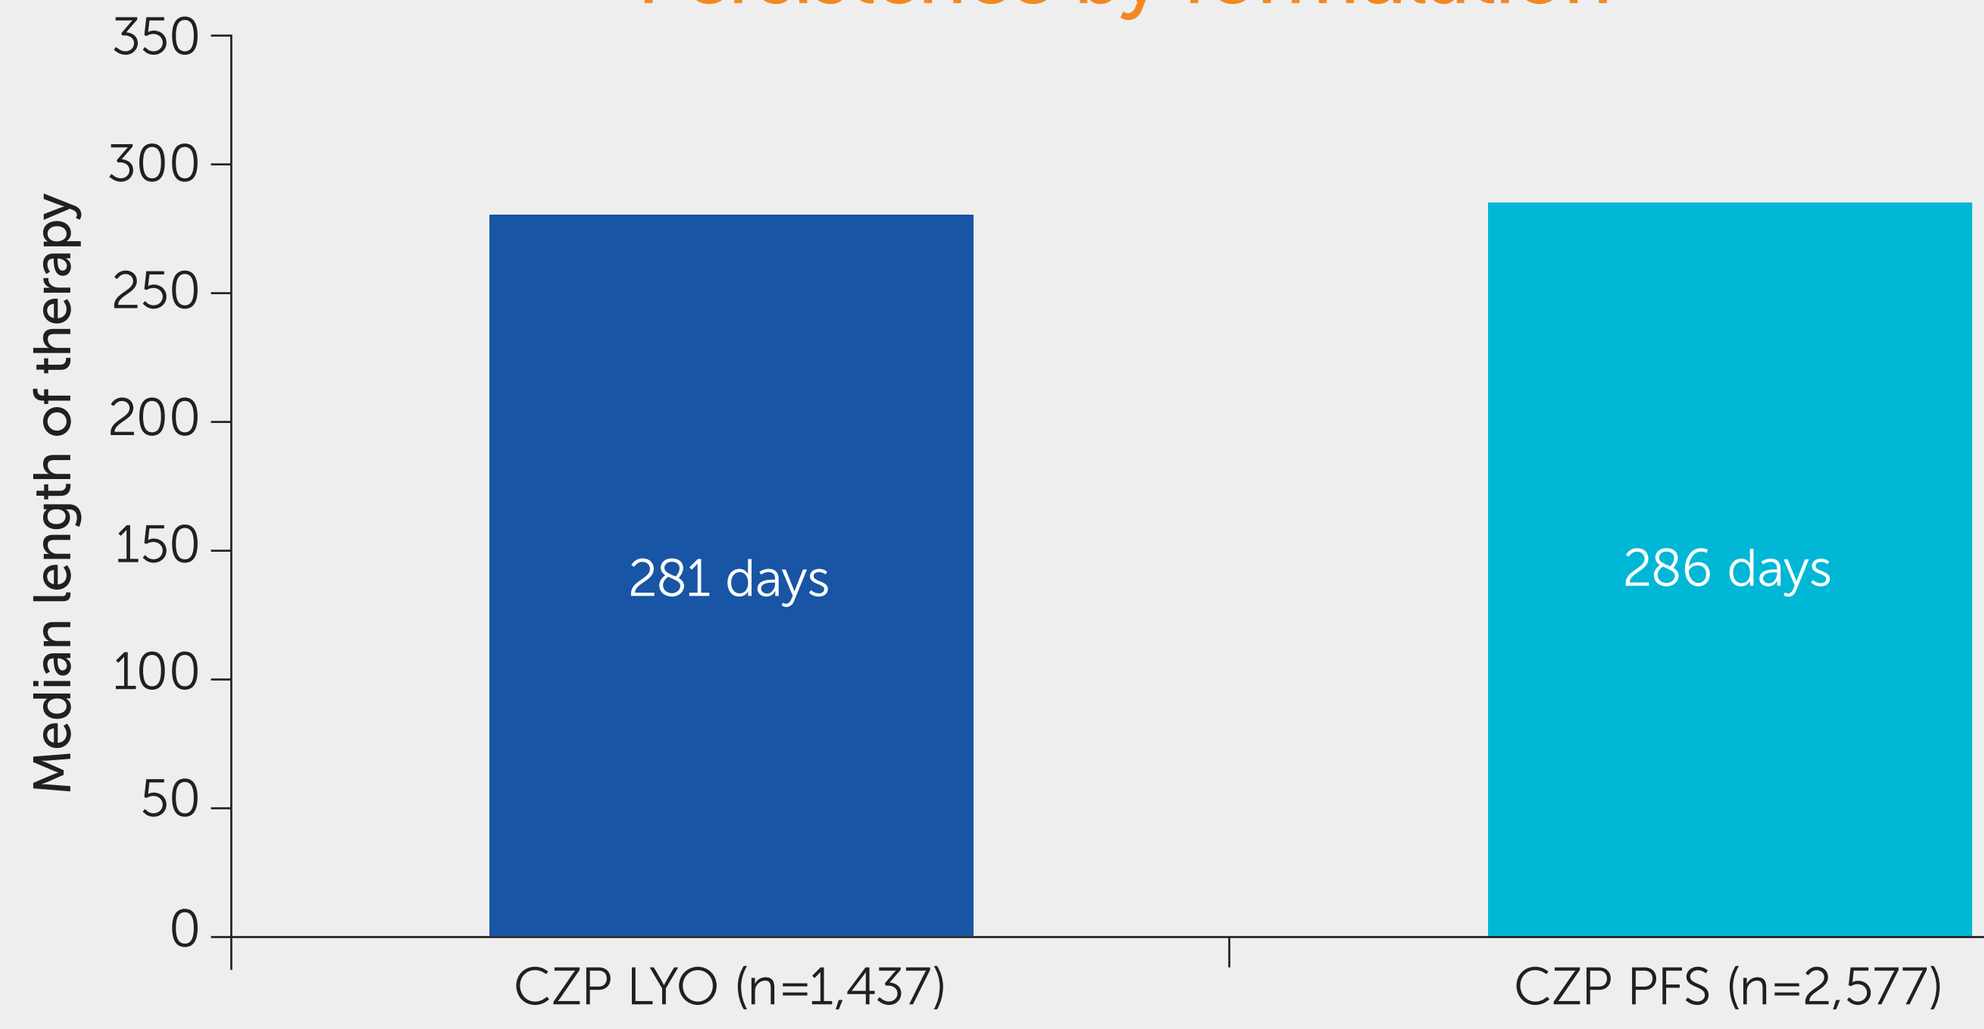
Median length of therapy
350
300
250
200
150
100
50
0
281 days
286 days
CZP LYO (n=1,437)
CZP PFS (n=2,577)

Event = discontinuation, formulation switch, or end of eligibility.

Adherence by formulation

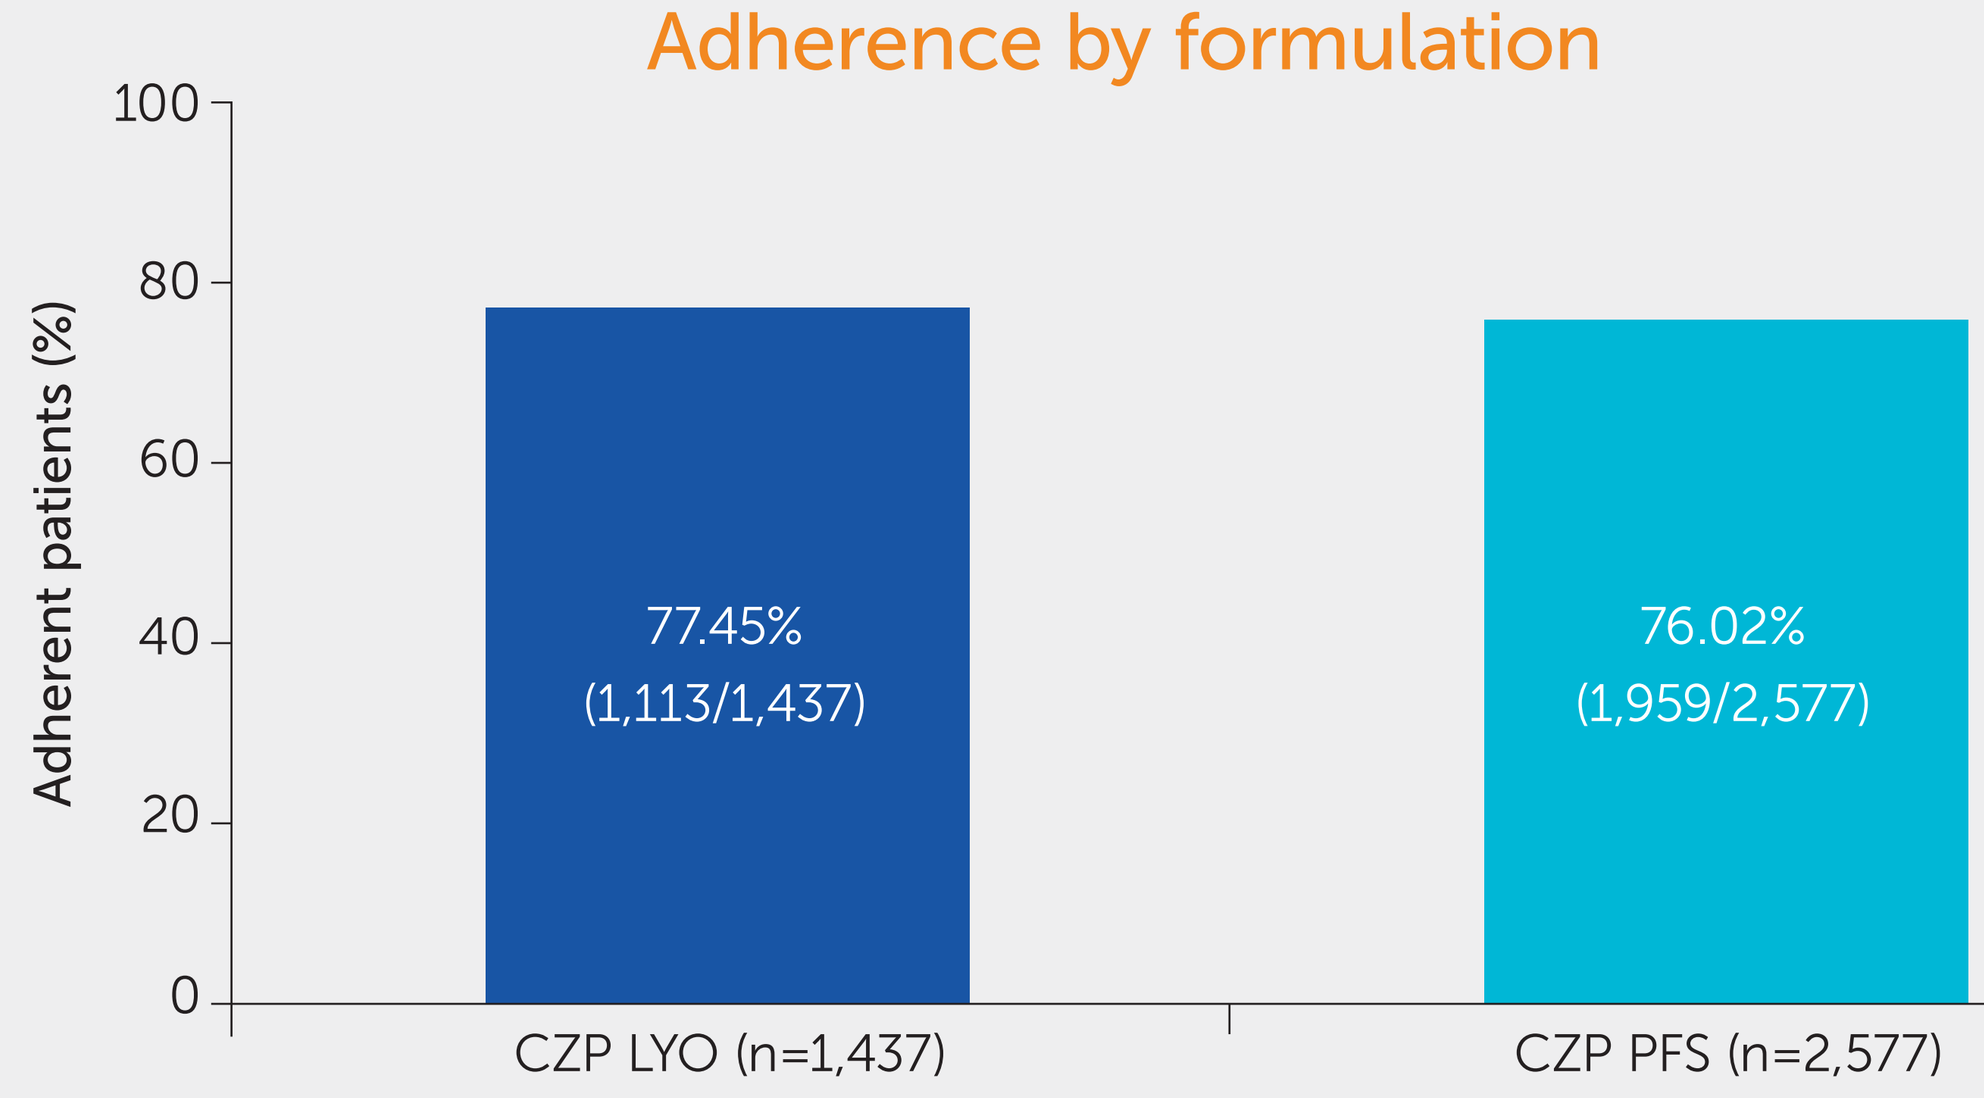
Adherent patients (%)
100
80
60
40
20
0
77.45%
(1,113/1,437)
76.02%
(1,959/2,577)
CZP LYO (n=1,437)
CZP PFS (n=2,577)

Adherence cut-off = 80%.

Initiation of CZP LYO was associated with older age, female sex, fewer baseline biologic treatments, and prior in-office biologic use

Figure 1 Study flow

Patients with ≥1 prescription claim for CZP
The index date was defined as the first CZP prescription
N=13,464

↓

Patients with ≥1 claim with a diagnosis of RA, PsA, or axSpA in the 12 months prior to the index date
N=7,816 (58.1%)

↓

Patients ≥18 years of age as of the index date
N=7,800 (99.8%)

↓

Patients with complete data availability (continous eligibility) during the 12 months prior to the index date (baseline period; 12 month washout period for CZP)
N=4,016 (51.5%)[a]

CZP LYO Cohort: Patients with an LYO formulation of CZP on the index date
n=1,437 (35.8%)

- ≥1 claim with a diagnosis of RA in the 12 months prior to the index date n=1,033 (71.9%)
- ≥1 claim with a diagnosis of PsA in the 12 months prior to the index date n=430 (29.9%)
- ≥1 claim with a diagnosis of axSpA in the 12 months prior to the index date n=264 (18.4%)

CZP PFS Cohort: Patients with a PFS formulation of CZP on the index date
n=2,577 (64.2%)

- ≥1 claim with a diagnosis of RA in the 12 months prior to the index date n=1,624 (63.0%)
- ≥1 claim with a diagnosis of PsA in the 12 months prior to the index date n=836 (32.4%)
- ≥1 claim with a diagnosis of axSpA in the 12 months prior to the index date n=643 (25.0%)

[a]Two patients with both formulations for CZP on the index date were excluded from the analysis.

Table 1 Baseline demographics and characteristics

	Total (N=4,014)	CZP Cohort LYO (n=1,437)	CZP Cohort PFS (n=2,577)	p-value
Age at index date				<0.001 *
Mean (SD)	50.23 (13.62)	55.33 (14.57)	47.39 (12.17)	
Region, n (%)				<0.001 *
Northeast	638 (15.89)	246 (17.12)	392 (15.21)	
North Central	579 (14.42)	149 (10.37)	430 (16.69)	
South	2,265 (56.43)	940 (65.41)	1,325 (51.42)	
West	528 (13.15)	102 (7.10)	426 (16.53)	
Unknown	4 (0.10)	0	4 (0.16)	
Health plan type, n (%)				0.7532
Capitated	423 (10.54)	148 (10.30)	275 (10.67)	
Non-capitated	3,591 (89.46)	1,289 (89.70)	2,302 (89.33)	
Sex, n (%)				0.2301
Male	927 (23.09)	316 (21.99)	611 (23.71)	
Female	3,087 (76.91)	1,121 (78.01)	1,966 (76.29)	
CCI				<0.001 *
Mean (SD)	0.57 (1.04)	0.72 (1.15)	0.49 (0.97)	
CCI comorbidities, n (%)				
Chronic pulmonary disease	744 (18.54)	302 (21.02)	442 (17.15)	<0.01 *
Diabetes without chronic complication	411 (10.24)	168 (11.69)	243 (9.43)	<0.05 *
Diabetes with chronic complication	179 (4.46)	93 (6.47)	86 (3.34)	<0.001 *
Peripheral vascular disease	168 (4.19)	110 (7.65)	58 (2.25)	<0.001 *
Cerebrovascular disease	156 (3.89)	85 (5.92)	71 (2.76)	<0.001 *
Renal disease	147 (3.66)	73 (5.08)	74 (2.87)	<0.001 *
Malignancies	122 (3.04)	65 (4.52)	57 (2.21)	<0.001 *
Congestive heart failure	109 (2.72)	57 (3.97)	52 (2.02)	<0.001 *
Dementia	13 (0.32)	11 (0.77)	2 (0.08)	<0.001 *
Other comorbidities, n (%)				
Hypertension	1,604 (39.96)	719 (50.03)	885 (34.34)	<0.001 *
Hyperlipidemia	1,432 (35.68)	642 (44.68)	790 (30.66)	<0.001 *
Crohn's disease	212 (5.28)	40 (2.78)	172 (6.67)	<0.001 *

Means and standard deviations are shown for continuous characteristics, and statistical comparisons were conducted using t-tests; counts and percentages are shown for categorical characteristics, and statistical comparisons were conducted using chi-squared tests. *Indicates statistical significance.

Figure 2 Biologics received during baseline period prior to CZP initiation

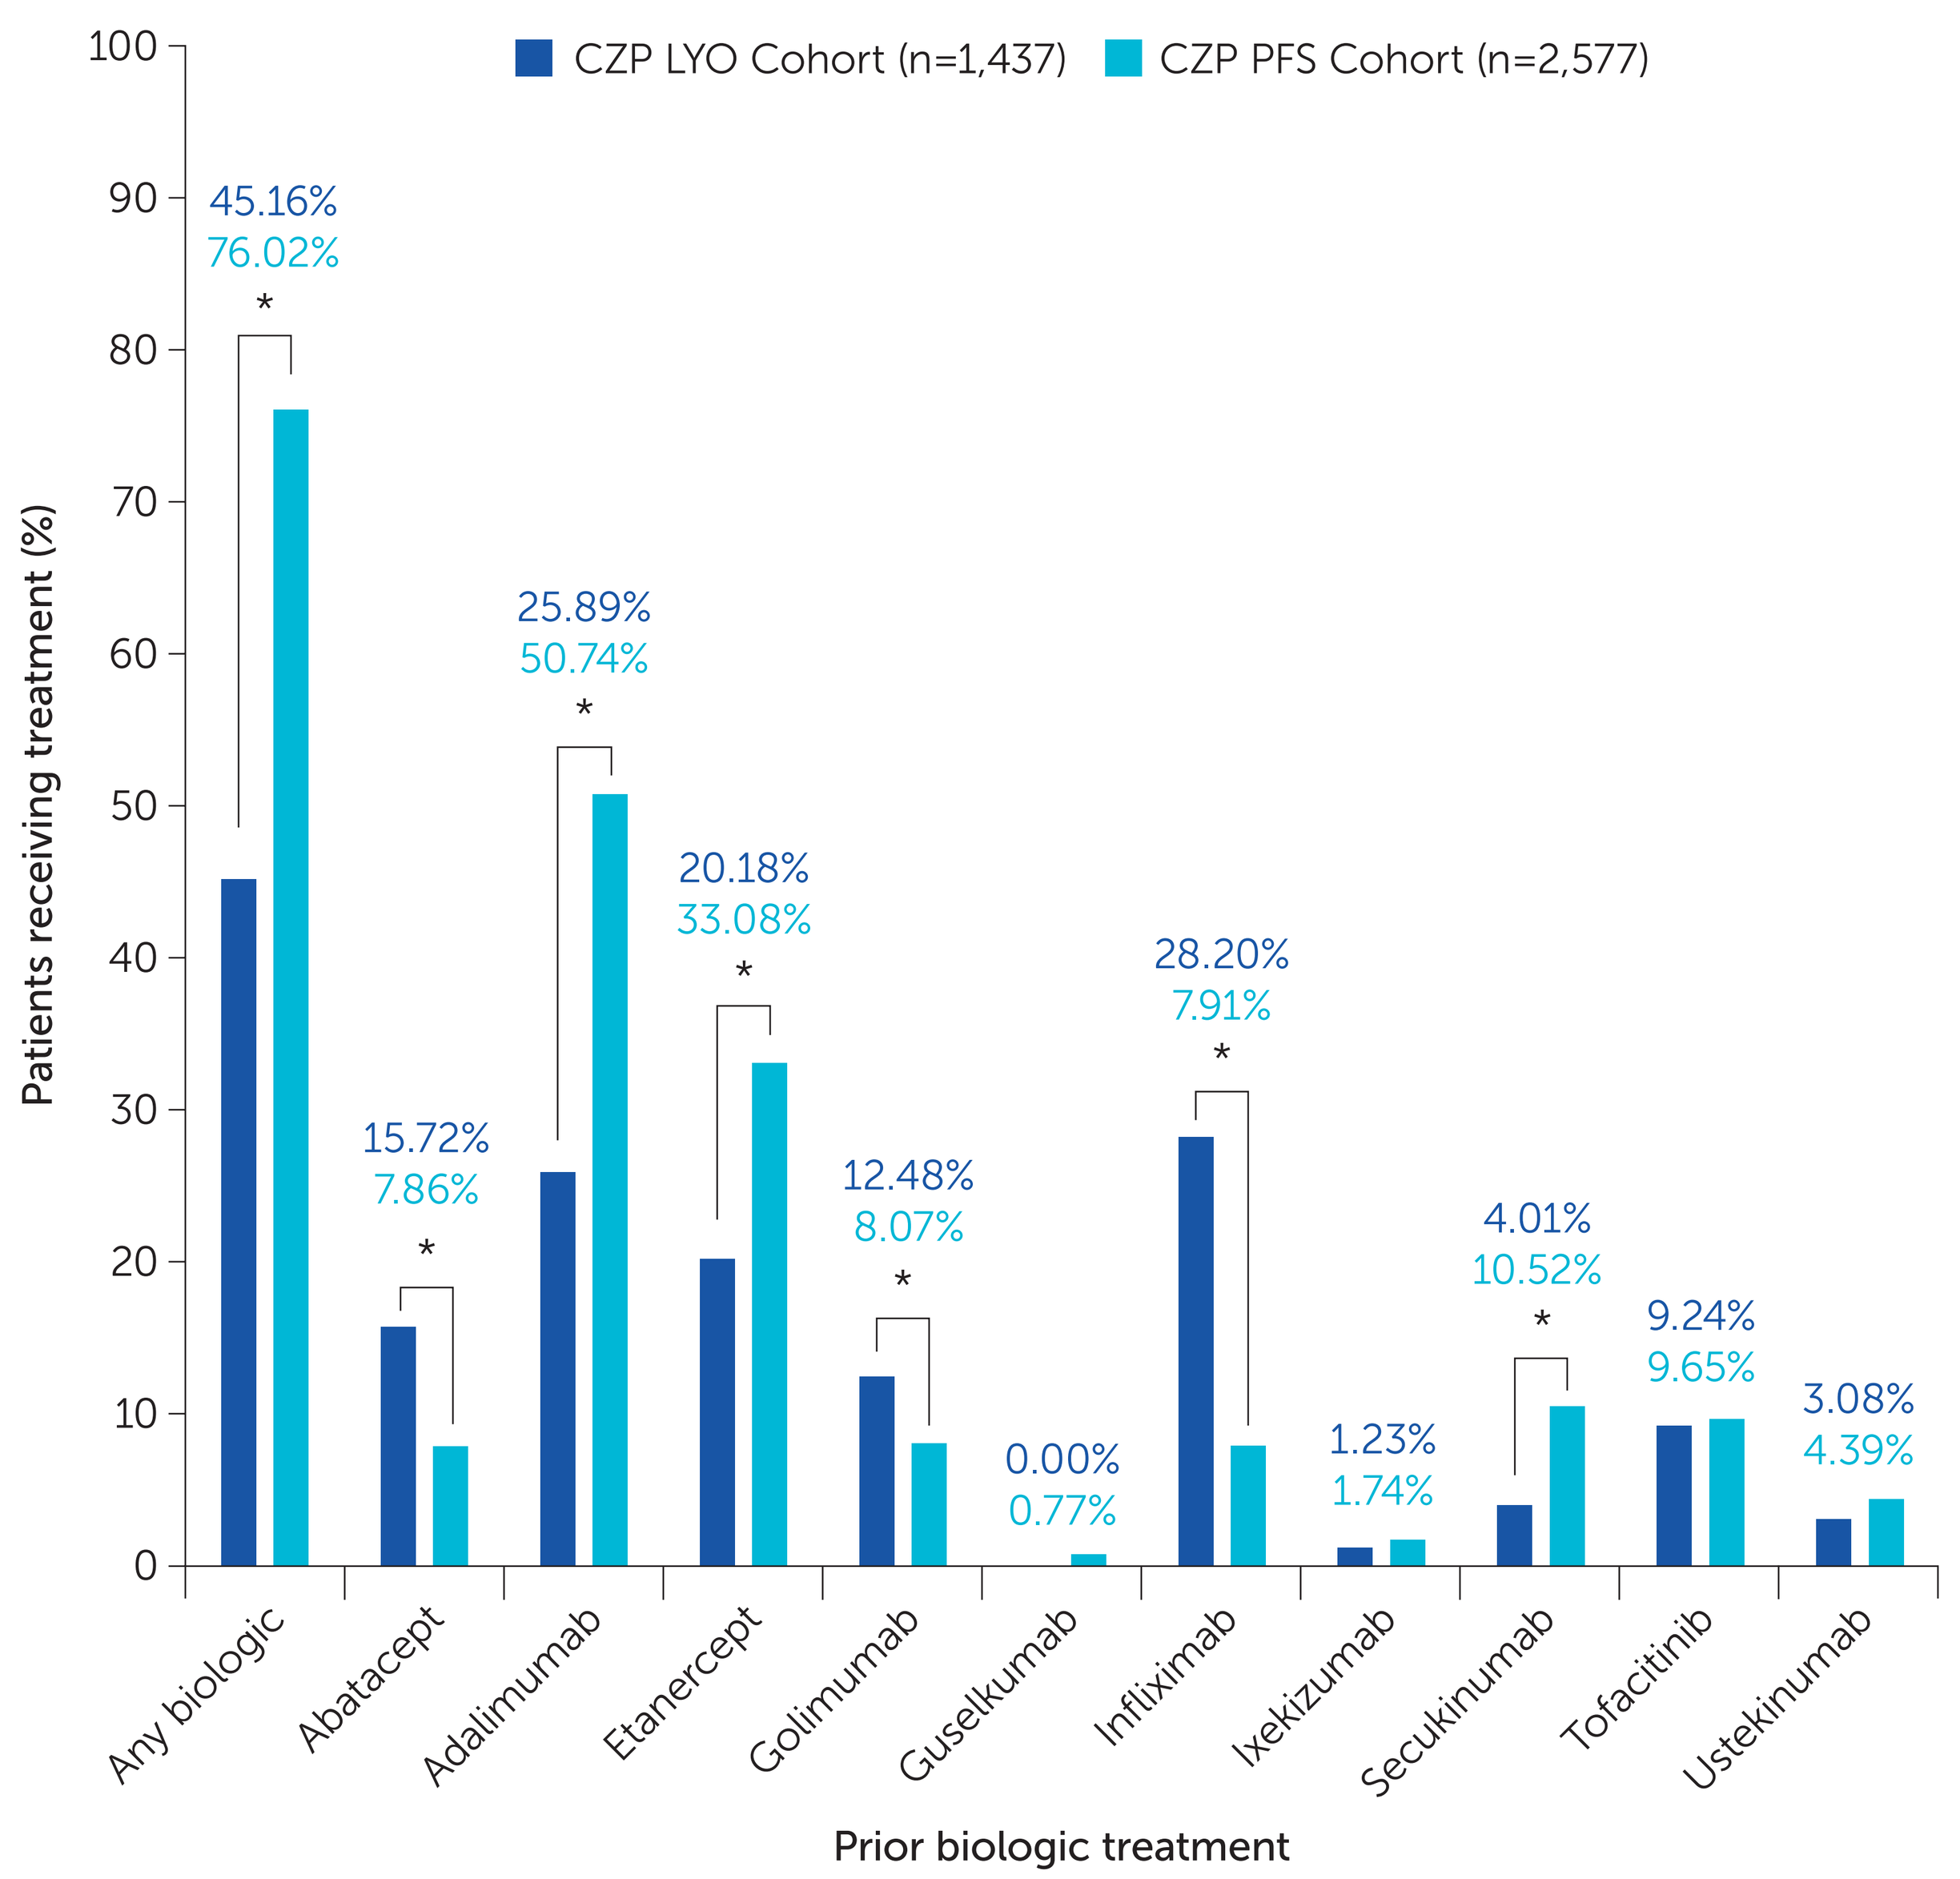
CZP LYO Cohort (n=1,437)
CZP PFS Cohort (n=2,577)
Patients receiving treatment (%)
100
90
80
70
60
50
40
30
20
10
0
45.16%
76.02%
15.72%
7.86%
25.89%
50.74%
20.18%
33.08%
12.48%
8.07%
0.00%
0.77%
28.20%
7.91%
1.23%
1.74%
4.01%
10.52%
9.24%
9.65%
3.08%
4.39%
Any biologic
Abatacept
Adalimumab
Etanercept
Golimumab
Guselkumab
Infliximab
Ixekizumab
Secukinumab
Tofacitinib
Ustekinumab
Prior biologic treatment

*p-value ≤0.05.

Table 2 Association of baseline characteristics with initiation of CZP LYO formulation†

Effect	Odds ratio	(95% CI)	p-value
Univariable analysis			
Baseline biologic adherence <80%	0.79	(0.65, 0.97) *	0.02 *
Multivariable analysis			
Baseline biologic adherence <80%	1.11	(0.88, 1.39)	0.37
Older age at index date	1.04	(1.03, 1.05)	<0.001 *
Male sex	0.73	(0.57, 0.92)	<0.01 *
Higher Charlson comorbidity index (CCI)	1.05	(0.96, 1.16)	0.28
Higher number of biologics used throughout baseline period	0.65	(0.52, 0.80)	<0.001 *
Last baseline biologic administration in home setting	0.12	(0.09, 0.15)	<0.001 *

†Logistic regression among biologic experienced patients; odds ratios for LYO vs. PFS initiation, 95% CIs, and p-values were estimated using a generalized linear model with a binomial distribution and a logit link function. *indicates statistical significance.

Figure 3 Adherence to biologic received prior to CZP

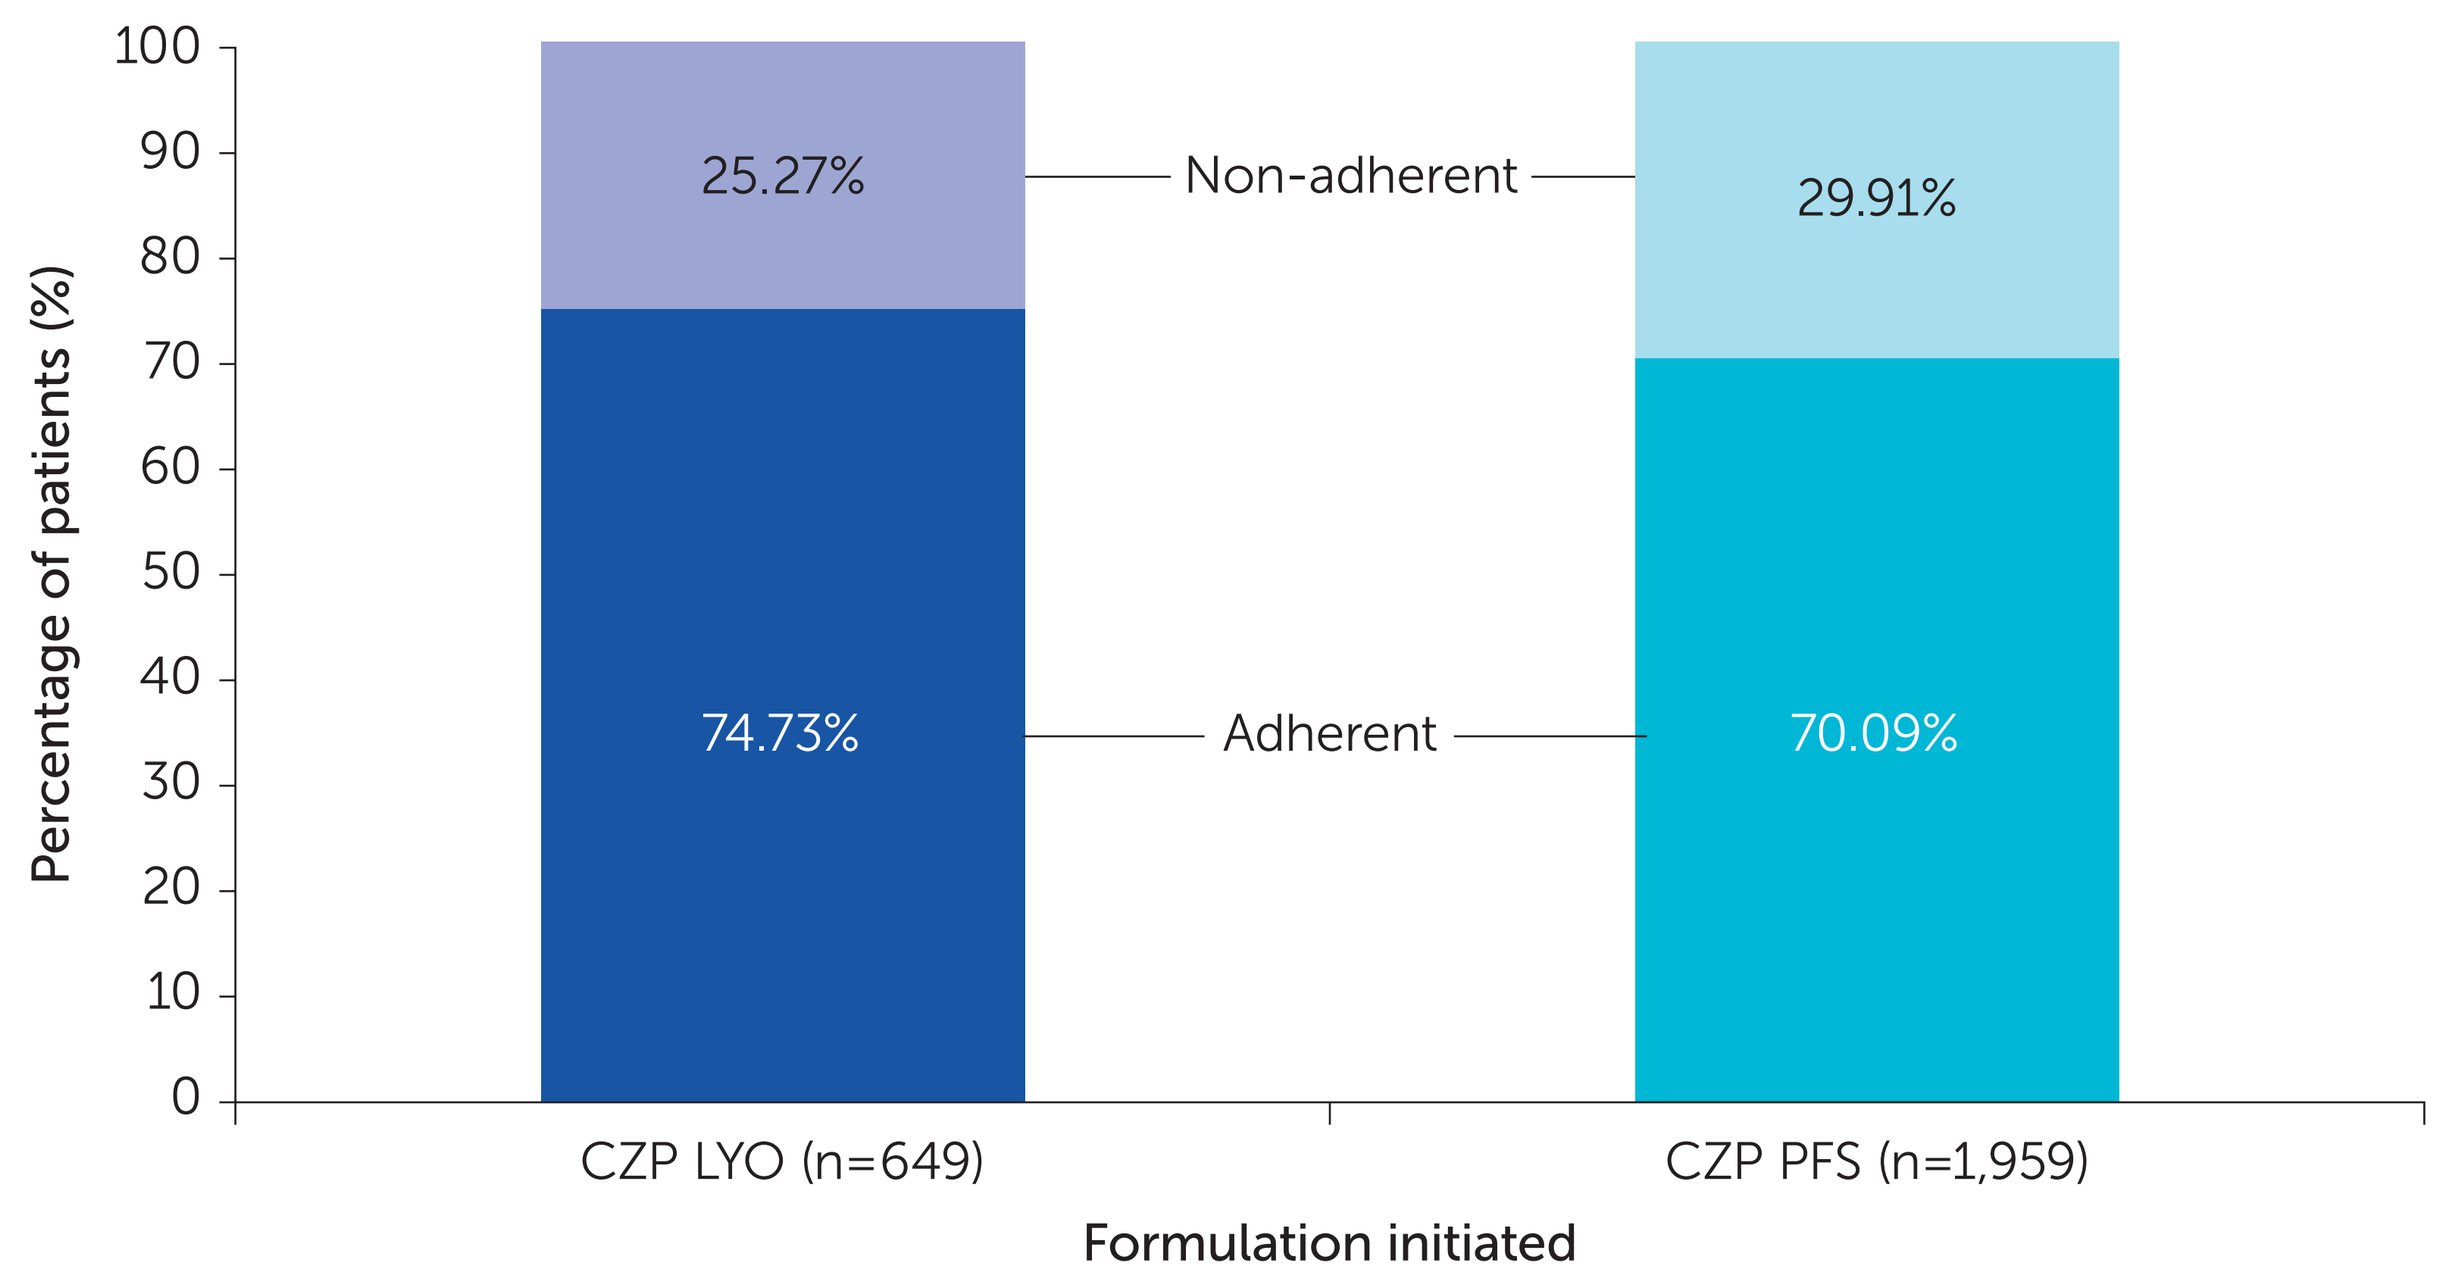
Percentage of patients (%)
100
90
80
70
60
50
40
30
20
10
0
25.27%
Non-adherent
29.91%
74.73%
Adherent
70.09%
CZP LYO (n=649)
CZP PFS (n=1,959)
Formulation initiated

Adherence cut-off = 80%.

Figure 4 Administration location of biologic prior to CZP

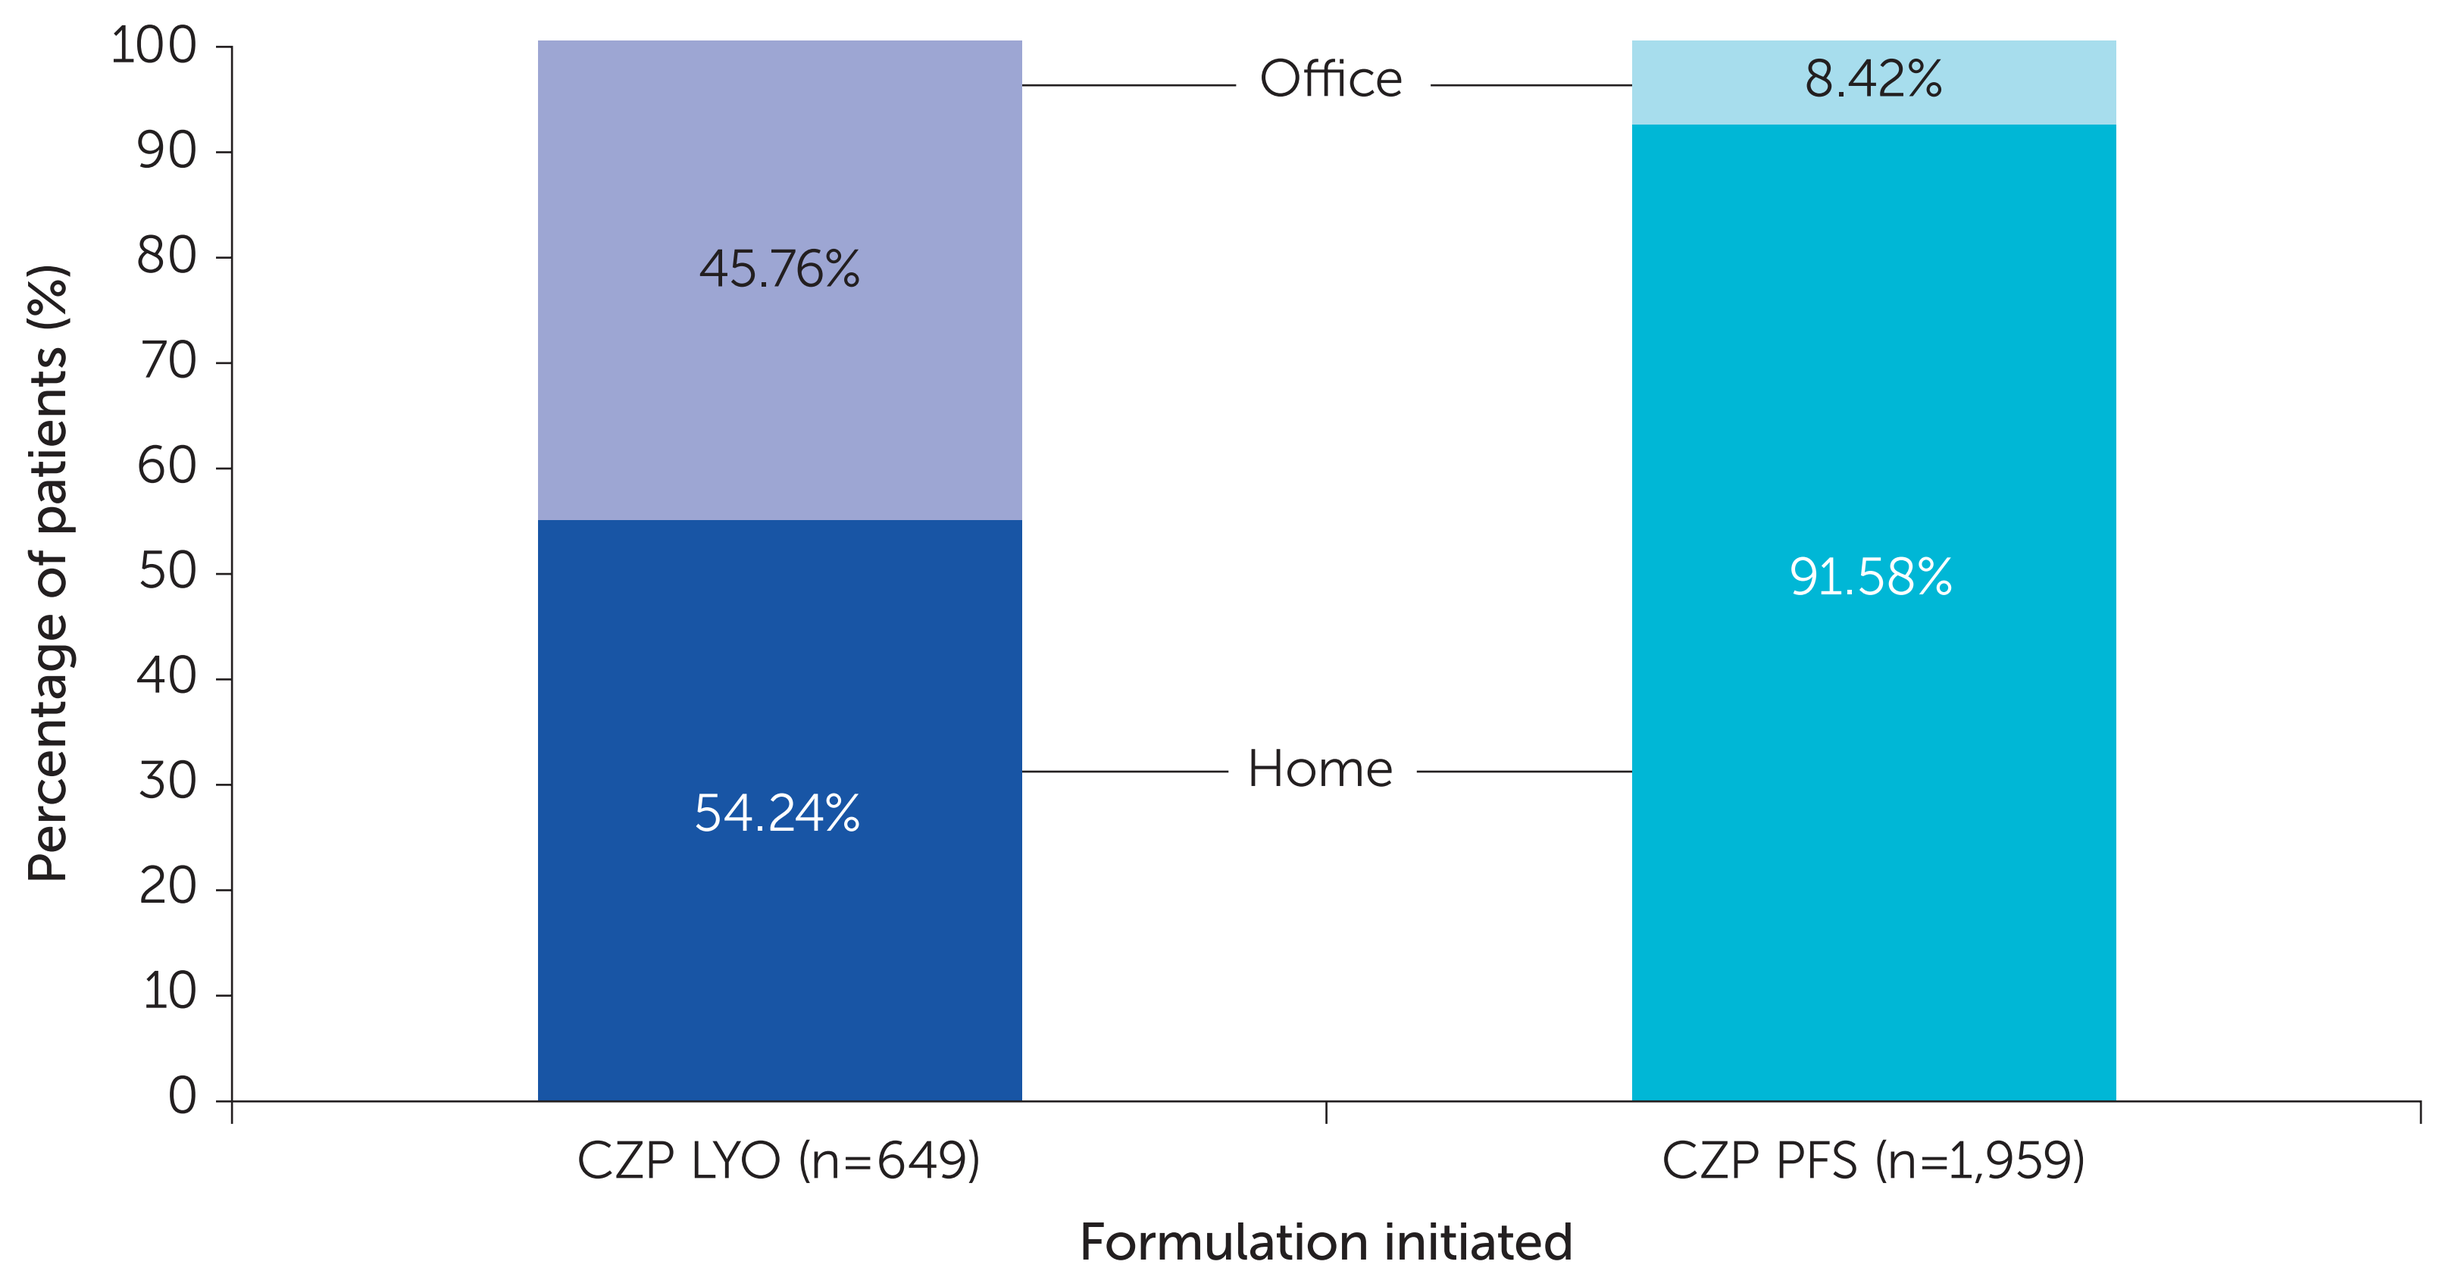
Percentage of patients (%)
100
90
80
70
60
50
40
30
20
10
0
Office
8.42%
45.76%
91.58%
Home
54.24%
CZP LYO (n=649)
CZP PFS (n=1,959)
Formulation initiated

Conclusions

Most patients who initiated CZP for axSpA, PsA, or RA, had high adherence and persistence, regardless of formulation.

Patients who initiated CZP LYO were older, had more comorbidities, and were more likely to be biologic-naïve compared to patients who initiated CZP PFS. In addition, patients who initiated CZP LYO were more likely to have received their last biologic immediately prior to CZP initiation in-office compared to patients who initiated CZP PFS.

AIDS: acquired immunodeficiency syndrome; axSpA: axial spondyloarthritis; CCI: Charlson Comorbidity Index; CZP: certolizumab pegol; HIV: human immunodeficiency virus; IQR: interquartile range; LYO: lyophilized powder; PFS: prefilled syringe; PsA: psoriatic arthritis; RA: rheumatoid arthritis; SD: standard deviation.

Institutions: [1]UCB Pharma, Smyrna, GA, USA; [2]Analysis Group, Inc., Boston, MA, USA; [3]Mercer University College of Pharmacy, Atlanta, GA, USA; *current affiliation: Bristol Myers Squibb, Lawrenceville, NJ, USA

References: [1]Raghunath S. *et al.* Clin Rheumatol. 2019;38(10):2699–2707; [2]Stolshek BS. *et al.* Am J Manag Care 2018; 24(8 Spec No.):SP315–SP321. Author Disclosures: **SB**: Employee and stockholder of UCB Pharma; **MLZ**, **FD**, **SP**, and **ES**: Employees of Analysis Group, Inc., which has received funding from UCB Pharma for the conduct of this study; **SH**: Employee of Bristol Myers Squibb, former employee of UCB Pharma; **JM**: Former intern of UCB Pharma. Acknowledgements: This study was funded by UCB Pharma. We would like to thank the patients and their caregivers in addition to all the investigators and their teams who contributed to this study. The authors acknowledge Mylene Serna, PharmD, UCB Pharma, Smyrna, GA, USA, for publication coordination, and Hannah L. Fox, PhD, Costello Medical, Boston, MA, USA, for medical writing support and editorial assistance. All costs associated with development of this presentation were funded by UCB Pharma.