Incidence of *De Novo* Depression Following Traumatic Brain Injury is Highest within 1 Year Post-injury

Samantha Nishimura, Kunal Lodaya, Jonathan Lilley, Sibyl Munson, Fabian T.W. D'Souza

Boston Strategic Partners, Inc.

BACKGROUND

Traumatic brain injuries (TBI) can result in substantial cognitive disruptions.[1] Depression is one of the most common complications after TBI,[2] known to hinder patient recovery and increase healthcare costs and resource utilization.[3,4] While pre-injury depression is a documented risk factor for post-injury depression,[2] it is suspected that many cases of post-injury depression are *de novo* diagnoses[5]

OBJECTIVES

We aimed to assess the incidence of *de novo* depression for three years post-TBI

METHODS

- De-identified patient data from a U.S. electronic health record database (Cerner Real-World Data) was extracted
- We included adults (≥18 years old) who sustained a TBI for the first time in 2017 and had at least 1 documented visit to a healthcare professional within 1 year both before and after injury
- We excluded those who previously experienced another serious neurological event (e.g., stroke, brain tumor, or neurodegenerative disease), had been diagnosed with depression, or used anxiolytic/antidepressant medications
- Cases of *de novo* depression were identified by either a diagnosis of a major depressive disorder (MDD) via ICD-10 codes (**Table 1**) or the receipt of an antidepressant or anxiolytic medication

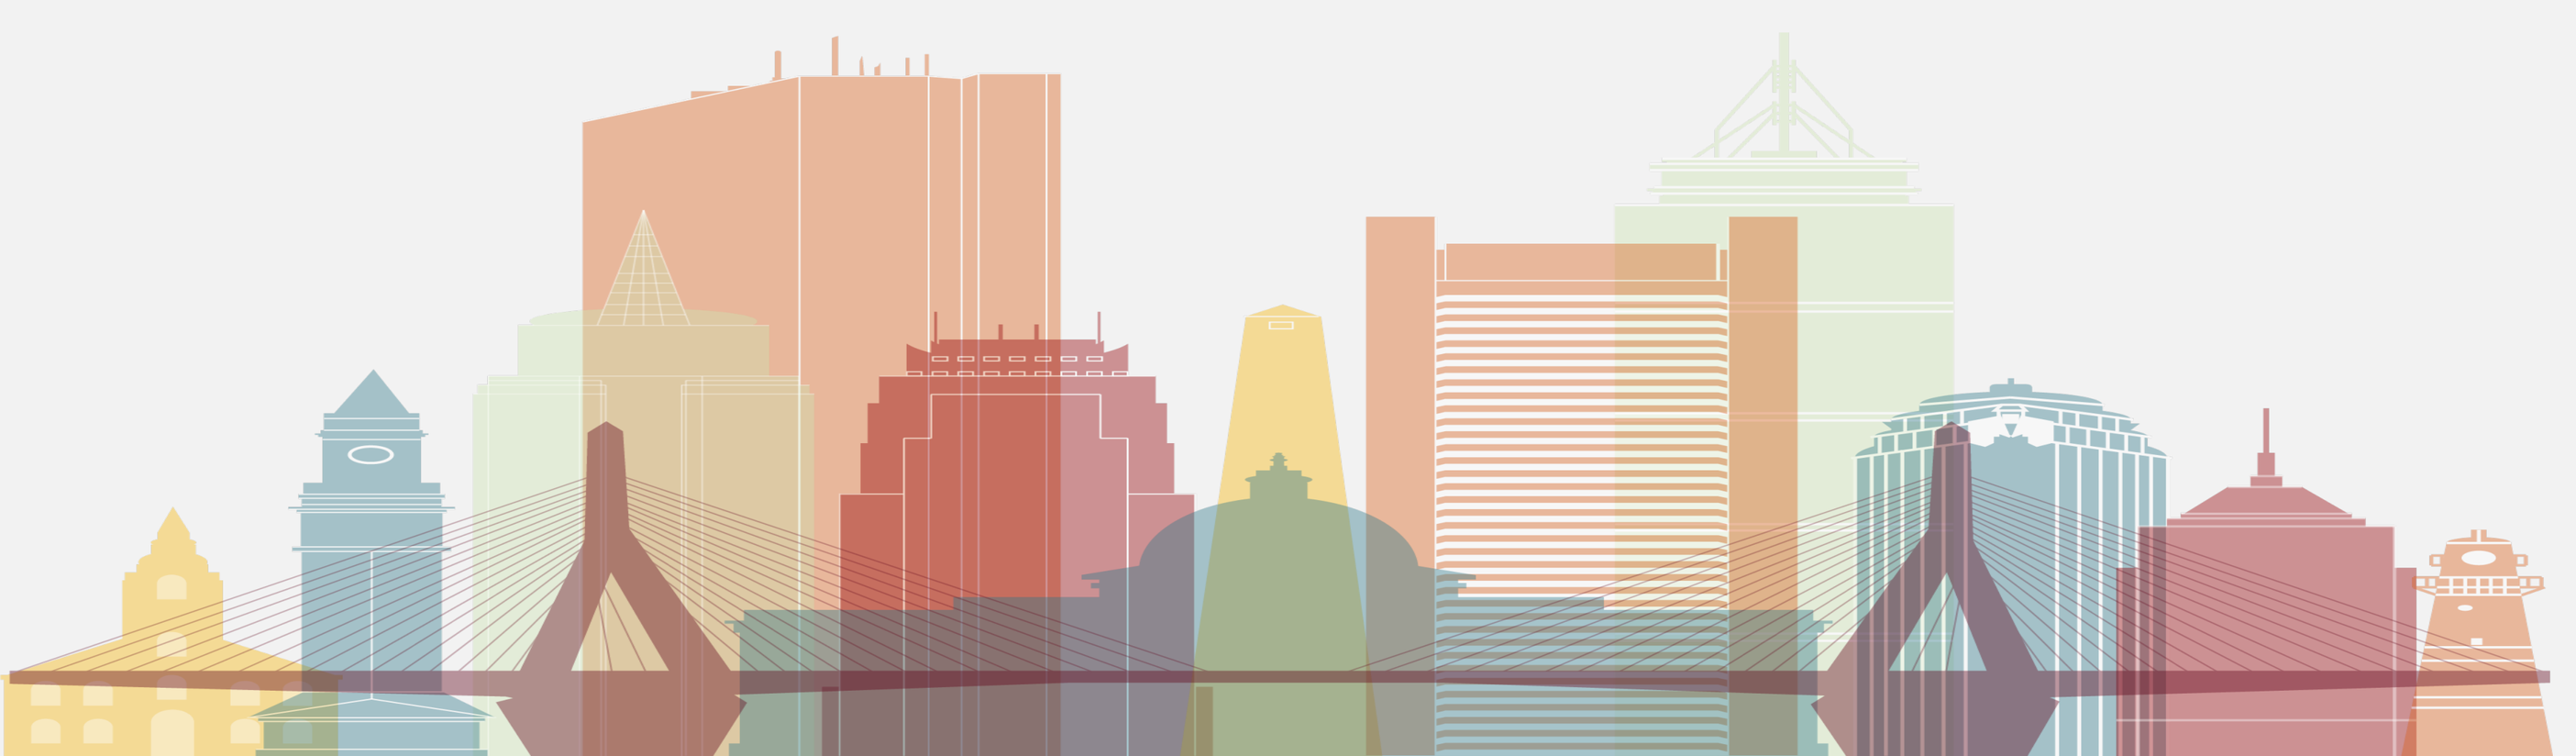

RESULTS

- A total of 10,399 patients experienced a TBI in 2017, of whom 52% were female
- The median age of patients who sustained a TBI was 39 years, but injury was most prevalent among patients aged 60+ (43.4% of cases)
- By 3 years post-injury, 861 patients (8.3%) were identified as having *de novo* depression, accounting for 7.3% of all males and 9.2% of all females
- The median age of those identified as having post-TBI *de novo* depression was 45
- The incidence of *de novo* depression decreased with time following a TBI, with 71.3% diagnosed by the end of Year 1 post-injury, a further 19.0% by the end of Year 2, and 9.7% by the end of Year 3 (**Figure 1**)
- The rate of *de novo* depression was 7.3% in Year 1, 0.8% in Year 2, and 0.3% in Year 3 (**Figure 2**)

CONCLUSIONS

- Over two-thirds of patients who experienced a TBI in 2017 were identified as having *de novo* depression within 1-year post-TBI
- To ensure timely diagnosis, healthcare providers should consider closely monitoring patients for symptoms of depression following their injury
- Future studies can further assess patients with TBI for additional psychiatric symptoms to gain a more comprehensive understanding of the epidemiology of mental health disorders

TABLE 1. ICD-10 codes used to identify cases of depression

Diagnosis	**Code(s)**
Depressive episode	F32.0, F32.1, F32.2, F32.4, F32.5, F32.9
Recurrent major depressive disorder	F33.0, F33.1, F33.2, F33.40, F33.41, F33.42, F33.9
Dysthymic disorder	F34.1

FIGURE 1. Incidence and timing of post-TBI *de novo* depression

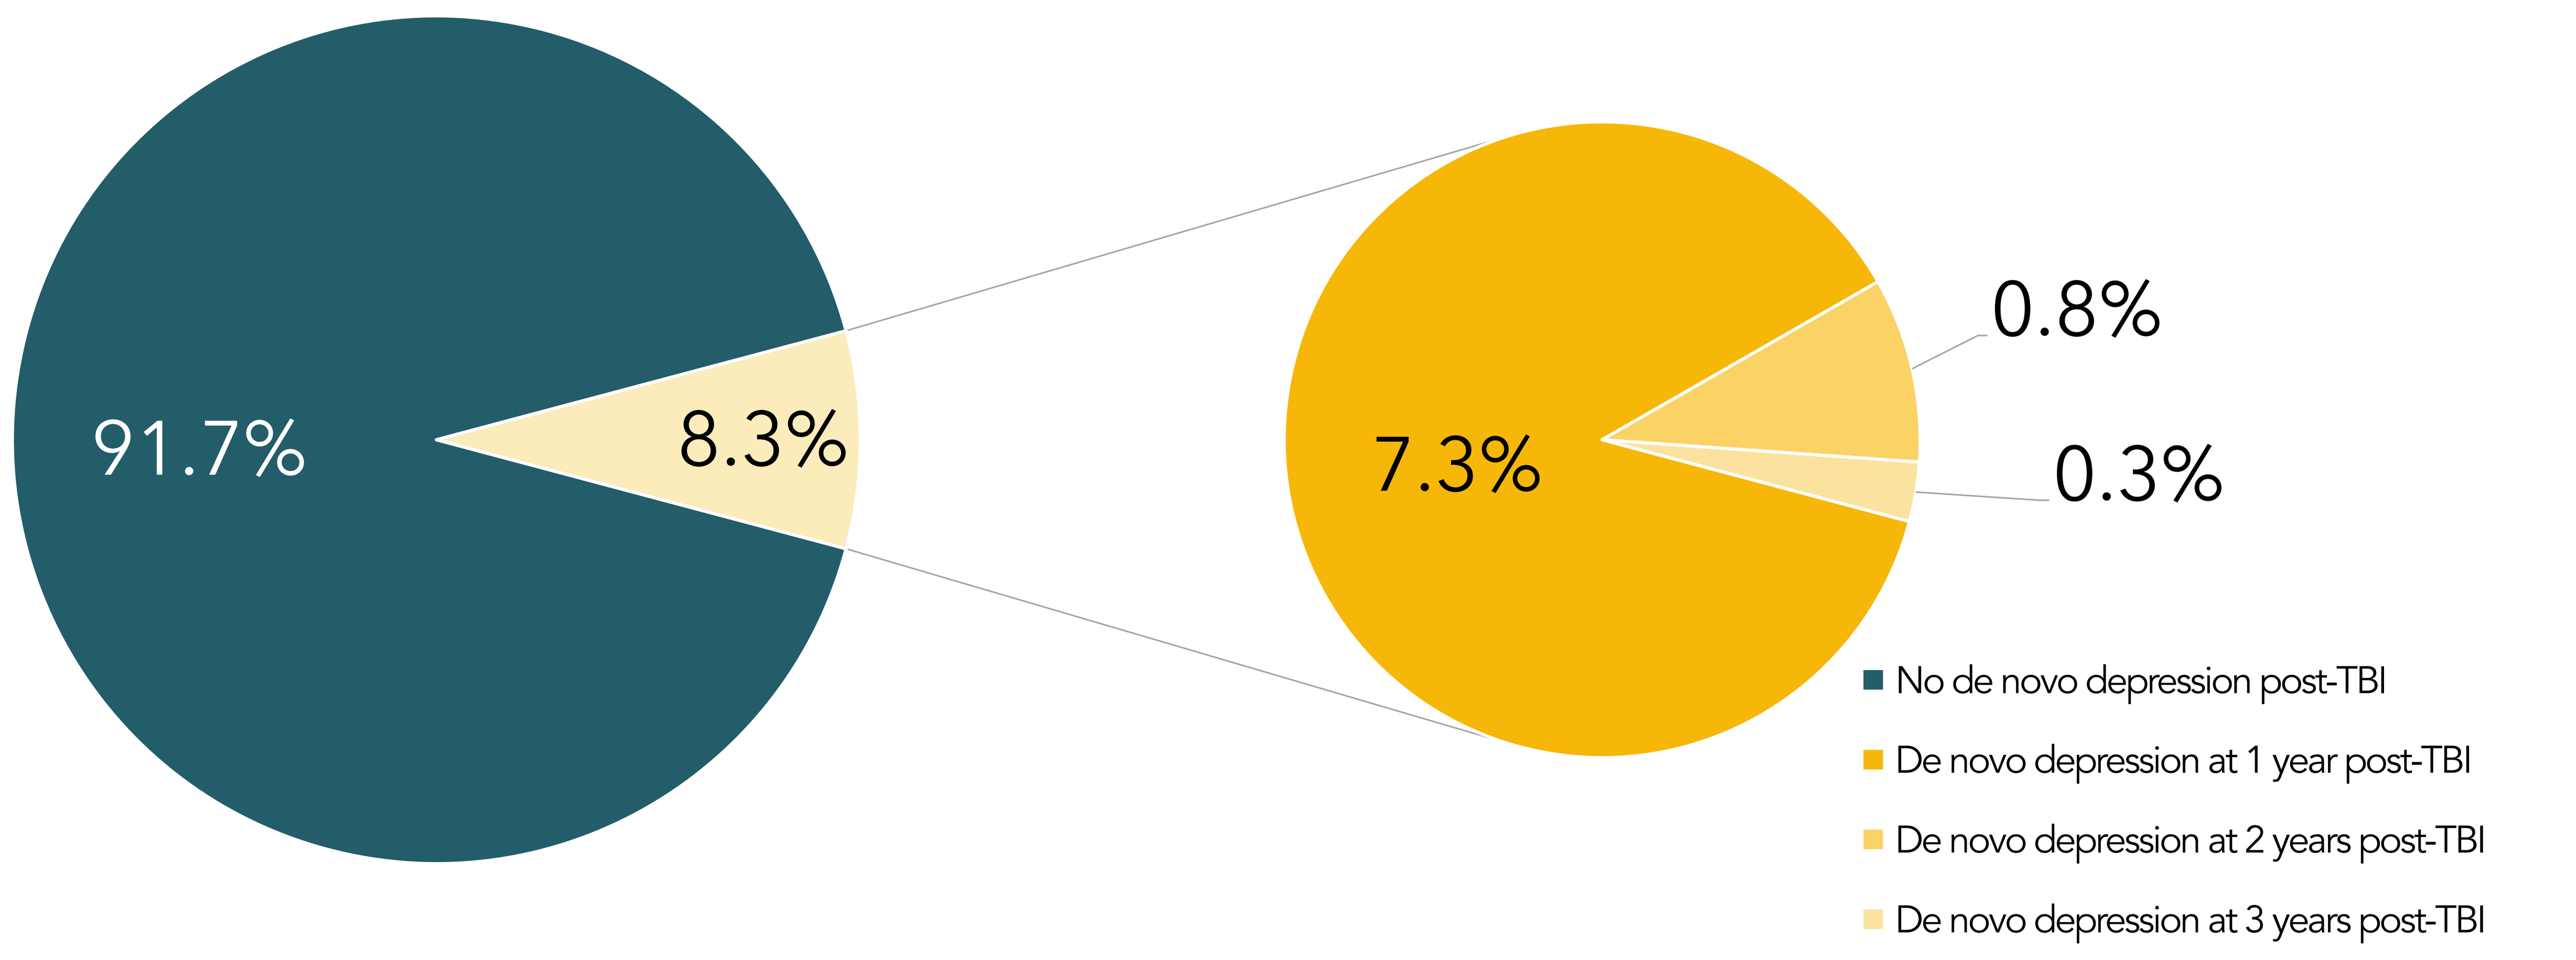
91.7%
8.3%
7.3%
0.8%
0.3%
No de novo depression post-TBI
De novo depression at 1 year post-TBI
De novo depression at 2 years post-TBI
De novo depression at 3 years post-TBI

FIGURE 2. Yearly incidence of *de novo* depression post-TBI

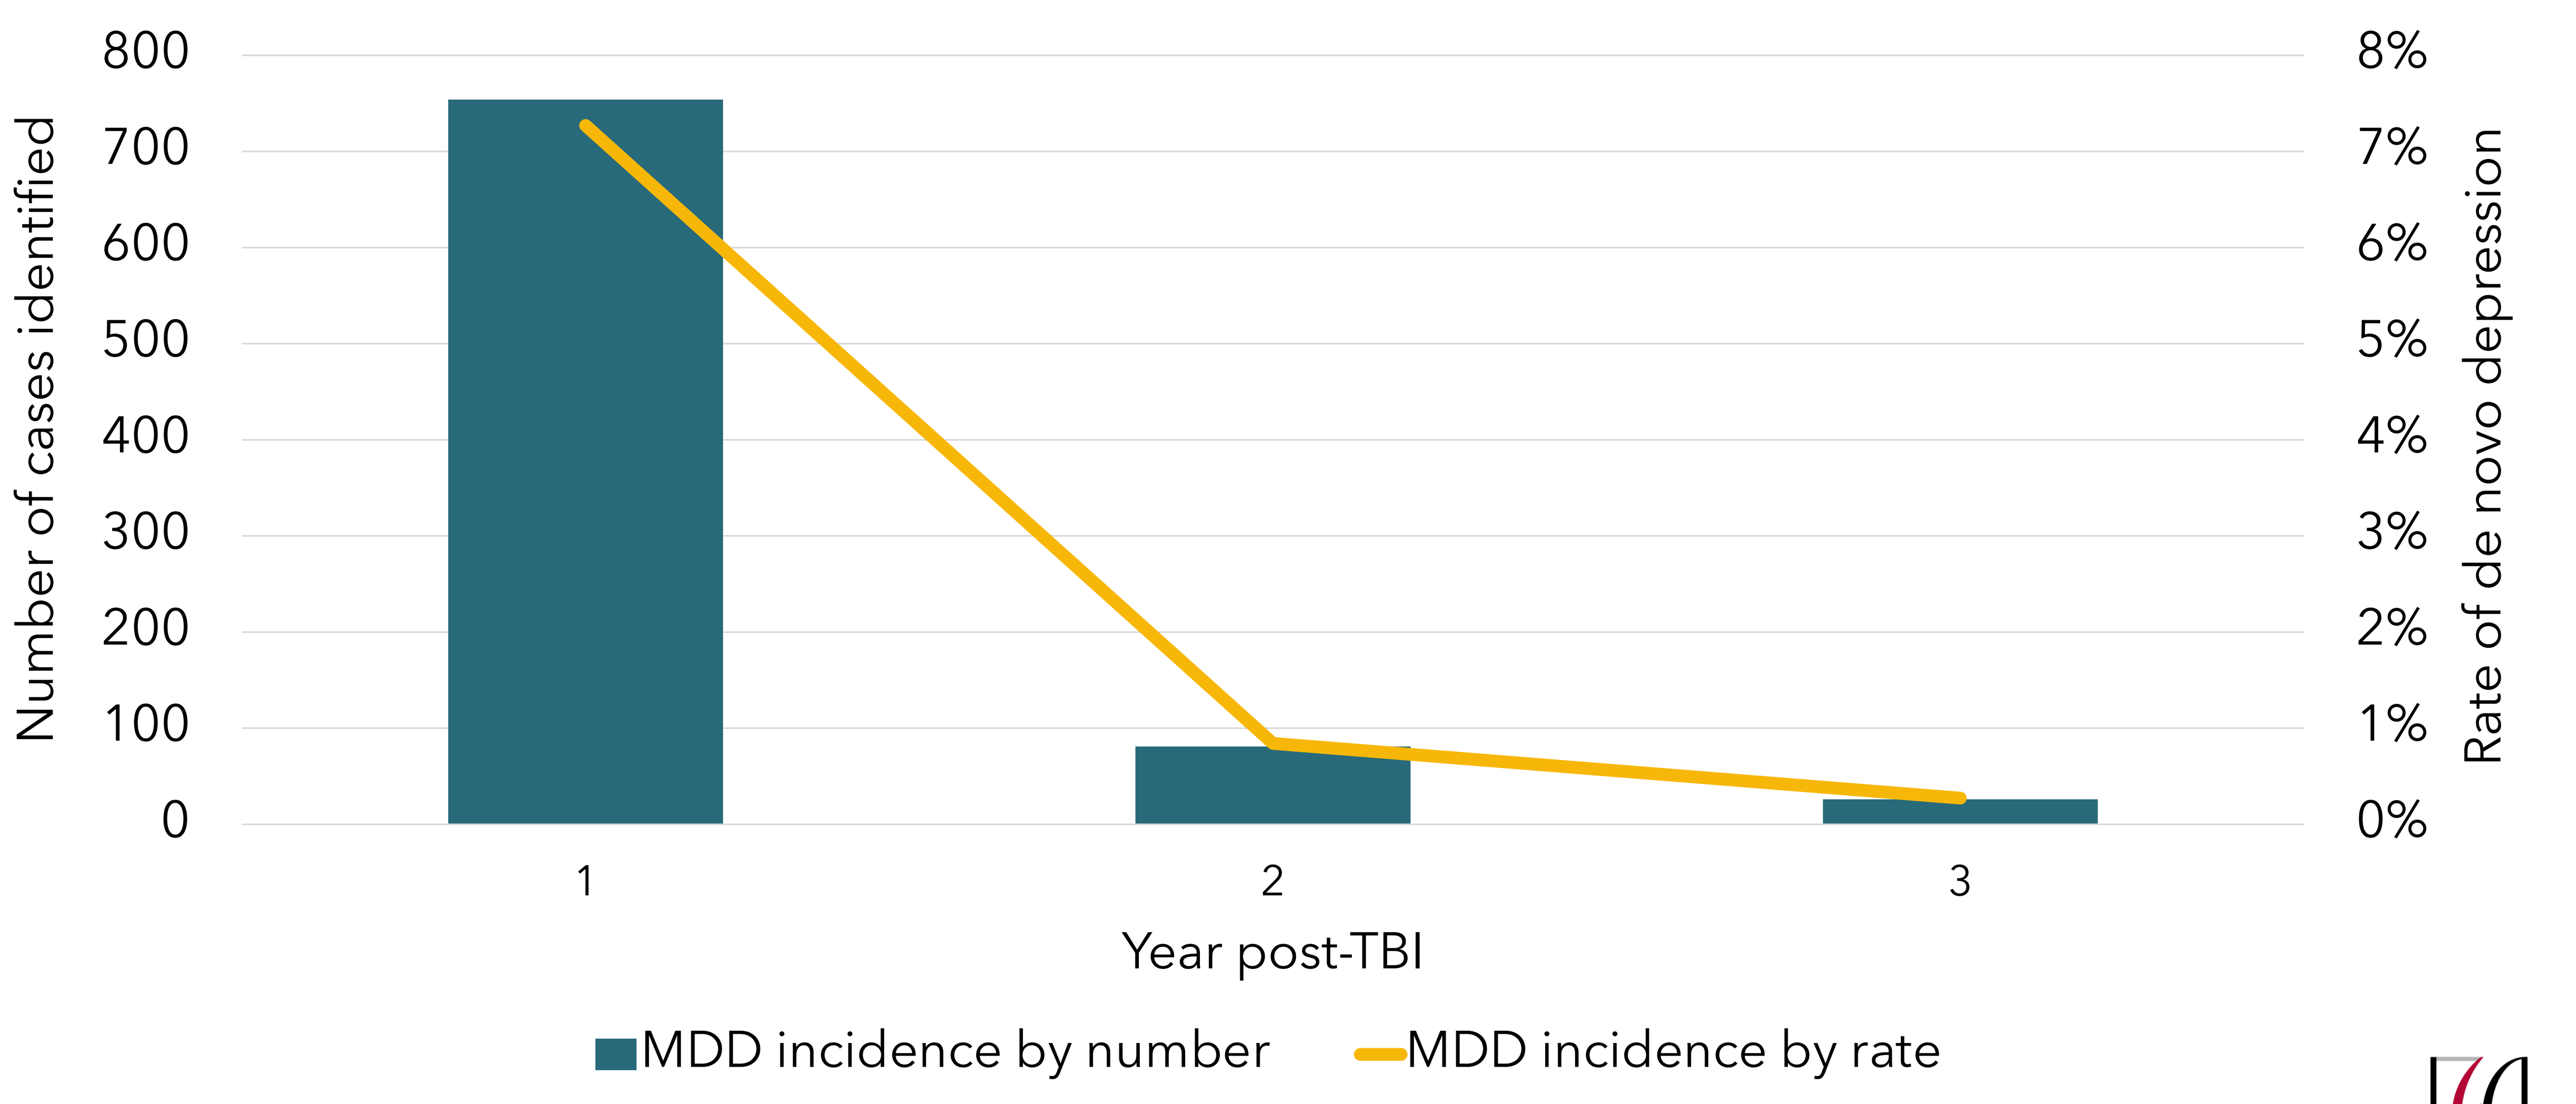
800
700
600
500
400
300
200
100
0
Number of cases identified
8%
7%
6%
5%
4%
3%
2%
1%
0%
Rate of de novo depression
1
2
3
Year post-TBI
MDD incidence by number
MDD incidence by rate

REFERENCES

1. Arciniegas, D. B., Held, K., & Wagner, P. (2002). Cognitive Impairment Following Traumatic Brain Injury. *Current Treatment Options in Neurology*, *4*(1), 43–57. https://doi.org/10.1007/s11940-002-0004-6. 2. Jorge, R. E., Robinson, R. G., Moser, D., Tateno, A., Crespo-Facorro, B., & Arndt, S. (2004). Major depression following traumatic brain injury. *Archives of General Psychiatry*, *61*(1), 42–50. https://doi.org/10.1001/archpsyc.61.1.42. 3. Gauthier, G., Guérin, A., Zhdanava, M., Jacobson, W., Nomikos, G., Merikle, E., François, C., & Perez, V. (2017). Treatment patterns, healthcare resource utilization, and costs following first-line antidepressant treatment in major depressive disorder: a retrospective US claims database analysis. *BMC Psychiatry*, *17*(1), 222. https://doi.org/10.1186/s12888-017-1385-0. 4. McIntyre, R. S., Prieto, R., Schepman, P., Yeh, Y.-C., Boucher, M., Shelbaya, A., Chambers, R., Gao, X., & Pappadopulos, E. (2019). Healthcare resource use and cost associated with timing of pharmacological treatment for major depressive disorder in the United States: a real-world study. *Current Medical Research and Opinion*, *35*(12), 2169–2177. https://doi.org/10.1080/03007995.2019.1652053. 5. Schachar, R. J., Park, L. S., & Dennis, M. (2015). Mental Health Implications of Traumatic Brain Injury (TBI) in Children and Youth. *Journal of the Canadian Academy of Child and Adolescent Psychiatry = Journal de l'Academie Canadienne de Psychiatrie de L'enfant et de L'adolescent*, *24*(2), 100–108. https://www.ncbi.nlm.nih.gov/pubmed/26379721.

EPH91