Associations Between Quality of Life, Major Depressive Disorder, Social Support, and Race/Ethnicity: A Moderation Analysis Using the 2021 US National Health and Wellness Survey

Kathryn L. Krupsky, Kushal Modi, Nate Way, M. Janelle Cambron-Mellott
Cerner Enviza, Malvern, PA, USA

PCR127

Introduction

- Major Depressive Disorder (MDD) is a serious mental health condition that affected 8.4% of adults in the US in 2020 (roughly 21 million adults).[1]
- Social support is one aspect of social environments associated with higher quality of life (QoL),[2] and among individuals with MDD, social support can buffer against serious negative outcomes associated with the condition.[3]
- Understanding the direct relationships between MDD and QoL, within the context of social support, may inform clinical practices seeking to improve QoL among patients with MDD.
- Whether social support can buffer the burden of MDD may also depend on an individual's societal context.[4]
- Examining potential race/ethnic differences in the relationships between MDD, QoL, and social support may generate essential evidence to inform clinical care among patients with MDD.

Objective

- The objective of this study was to examine whether social support and race/ethnicity moderates the association between MDD and QoL.

Methods

Data Source

- Data were from the 2021 US National Health and Wellness Survey (NHWS), an internet-based, self-reported survey designed to represent the general adult population, relative to age, gender, and race/ethnicity distributions reported in the US Census (N=75,098).

Key Variables

Variable	Operationalization
MDD Status (binary)	The Patient Health Questionnaire (PHQ-9) to screen for MDD (positive screen=PHQ-9 ≥10)
QoL (continuous)	Short-Form Six Dimensions utility index score (SF-6D Utility Index)
Social Support (continuous)	Modified Medical Outcomes Study Social Support Survey (mMOS-SS) overall summary score
Race/Ethnicity (categorical)	Self-reported race/ethnicity categorized as White, Black/African American, Asian, Hispanic, and 2+ races

Eligibility

- Inclusion criteria:
 - Eligible to participate in the US NHWS
 - Self-reported their race/ethnicity as either White, Black/African American, Asian, Hispanic, or 2+ races
- Exclusion criteria:
 - Self-reported ever experiencing bipolar depression or schizophrenia
 - Had missing data for any variable relevant to analytic strategy

Statistical Analyses

- Descriptive (means, standard deviations, percentages, sample sizes) and bivariate (Chi-square tests and one-way ANOVA) statistics were used to characterize the sample overall and according to MDD status.
- We used hierarchical (sequential) regression analyses to test the moderating effect of race/ethnicity and social support on the relationship between MDD and QoL. This involved creating three two-way interaction terms from the product of the main effect terms, as well as a three-way interaction term. We constructed our regression model in a stepwise fashion (**Figure 1**), and after each modeling step, we used the F-test, R^2, and ΔR^2 to compare predictive validity of each model. Given the three-way interaction was statistically significant (α=0.05), we probed the interaction by plotting estimated covariate-adjusted slopes and slope differences.

Methods, continued

Figure 1. Models Tested in Hierarchical (Sequential) Regression Analysis with the SF-6D Utility Index Score Modeled as the Outcome Variable Assuming a Normal Distribution

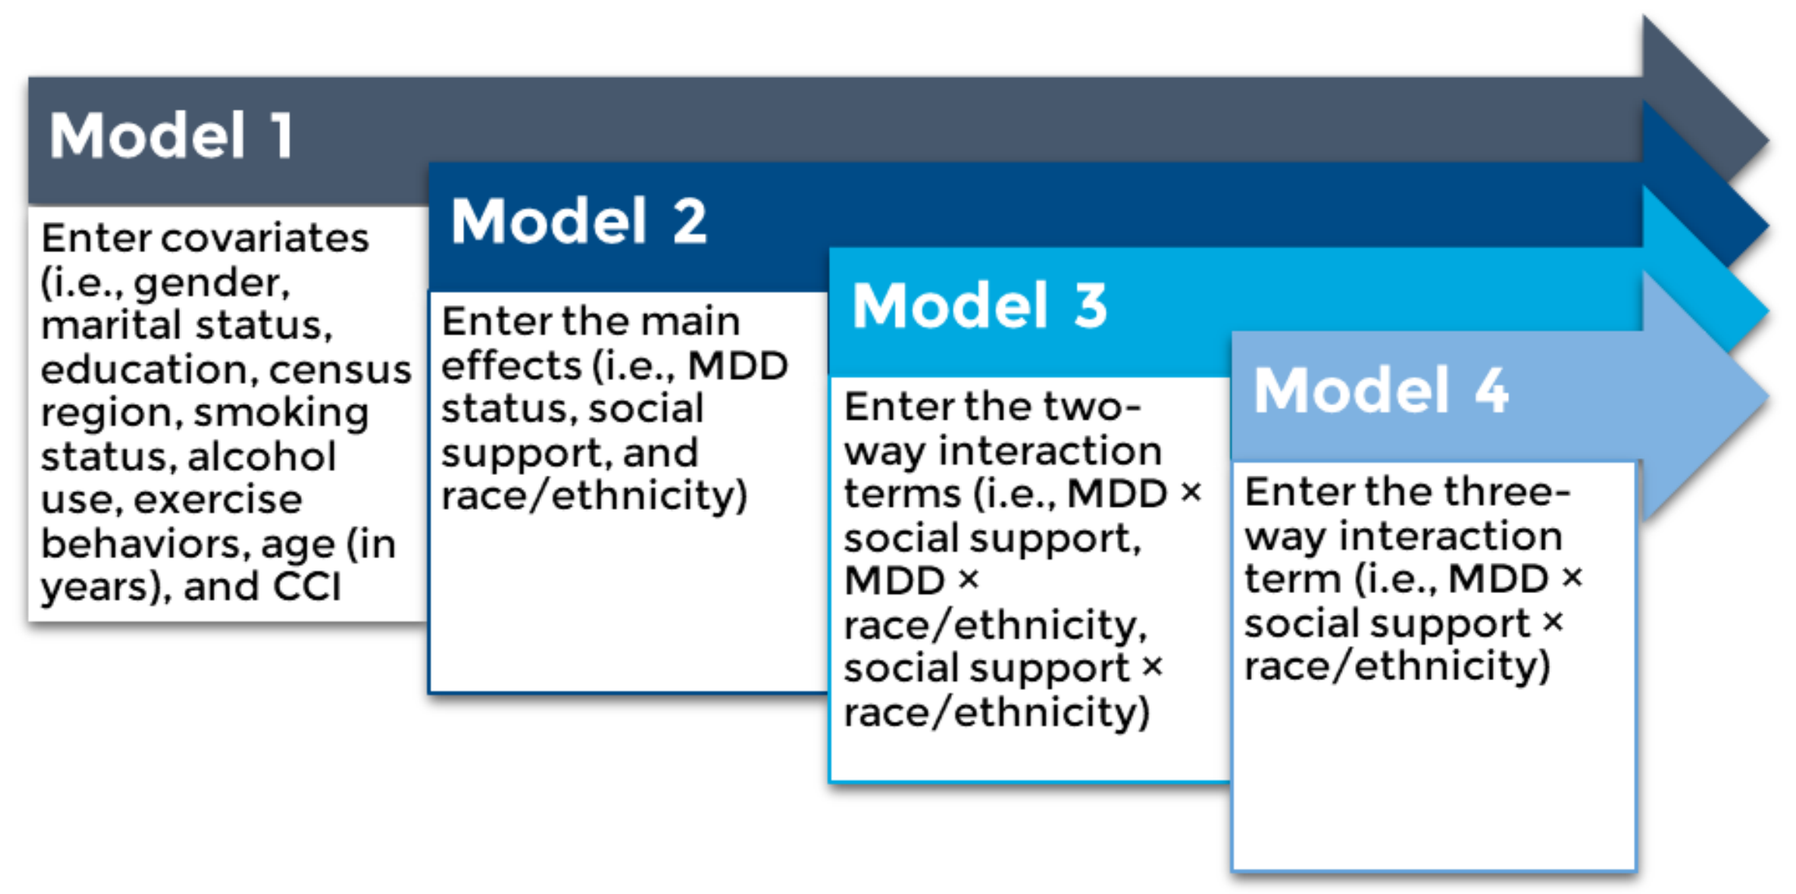
Model 1
Enter covariates (i.e., gender, marital status, education, census region, smoking status, alcohol use, exercise behaviors, age (in years), and CCI
Model 2
Enter the main effects (i.e., MDD status, social support, and race/ethnicity)
Model 3
Enter the two-way interaction terms (i.e., MDD × social support, MDD × race/ethnicity, social support × race/ethnicity)
Model 4
Enter the three-way interaction term (i.e., MDD × social support × race/ethnicity)

Results

Sample Characteristics

- Our analytic sample consisted of 69,279 respondents, of whom, 14,985 (21.6%) had MDD, while 54,294 (78.4%) did not have MDD.
 - Mean age=49.2 years; 56.2% female
 - 69.3% White, 12.5% Hispanic, 9.1% Black/African American, 6.9% Asian, 2.1% 2+ races (**Table 1**)
- Compared to adults without MDD, respondents with MDD were younger, less often married or living with a partner, less educated, had lower levels of household income, and represented by a higher proportion of Hispanic and Black/African American respondents (*p*-values <0.001) (**Table 1**).
- Individuals with MDD had significantly lower QoL and reported significantly lower levels of social support than individuals without MDD (0.56 vs. 0.76, p <0.001; 27.0 vs. 29.9, p <0.001, respectively) (**Figures 2 and 3**).

Table 1. Characteristics of Analytic Sample Overall and According to MDD Status

Characteristics	Overall Sample (N=69,279)	MDD Status: With MDD (n=14,985)	MDD Status: Without MDD (n=54,294)	*p-Value*
Age in Years (Mean ± SD)	49.2 ± 17.5	38.1 ± 13.6	52.3 ± 17.2	<0.001
Sex, n (%)				
Male	30,346 (43.8%)	6,607 (44.1%)	23,739 (43.7%)	0.420
Female	38,933 (56.2%)	8,378 (55.9%)	30,555 (56.3%)	
Marital Status, n (%)				
Married/living with a partner	42,625 (61.5%)	8,440 (56.3%)	34,185 (63.0%)	<0.001
Single, not married/divorced/separated/widowed	16,929 (24.4%)	4,975 (33.2%)	11,954 (22.0%)	
Declined to answer	9,725 (14.0%)	1,570 (10.5%)	8,155 (15.0%)	
Race/Ethnicity, n (%)				
White	48,007 (69.3%)	8,390 (56.0%)	39,617 (73.0%)	<0.001
Black/African American	6,332 (9.1%)	1,676 (11.2%)	4,656 (8.6%)	
Asian	4,803 (6.9%)	922 (6.2%)	3,881 (7.1%)	
Hispanic	8,687 (12.5%)	3,610 (24.1%)	5,077 (9.4%)	
2+ races	1,450 (2.1%)	387 (2.6%)	1,063 (2.0%)	
Educational Attainment, n (%)				
HS degree or less	11,256 (16.2%)	3,596 (24.0%)	7,660 (14.1%)	<0.001
Some college	19,573 (28.3%)	4,414 (29.5%)	15,159 (27.9%)	
Bachelor's degree	21,743 (31.4%)	3,640 (24.3%)	18,103 (33.3%)	
Graduate degree	16,707 (24.1%)	3,335 (22.3%)	13,372 (24.6%)	
Annual Household Income, n (%)				
<\$25,000	7,318 (10.6%)	2,308 (15.4%)	5,010 (9.2%)	<0.001
\$25,000 to <\$50,000	11,881 (17.1%)	2,644 (17.6%)	9,237 (17.0%)	
\$50,000 to <\$100,000	11,305 (16.3%)	2,030 (13.5%)	9,275 (17.1%)	
\$100,000+	35,065 (50.6%)	7,472 (49.9%)	27,593 (50.8%)	
Declined to answer	3,710 (5.4%)	531 (3.5%)	3,179 (5.9%)	

Results, continued

Figure 2. QoL was Significantly Lower among Individuals with MDD vs. Individuals without MDD (p <0.001)

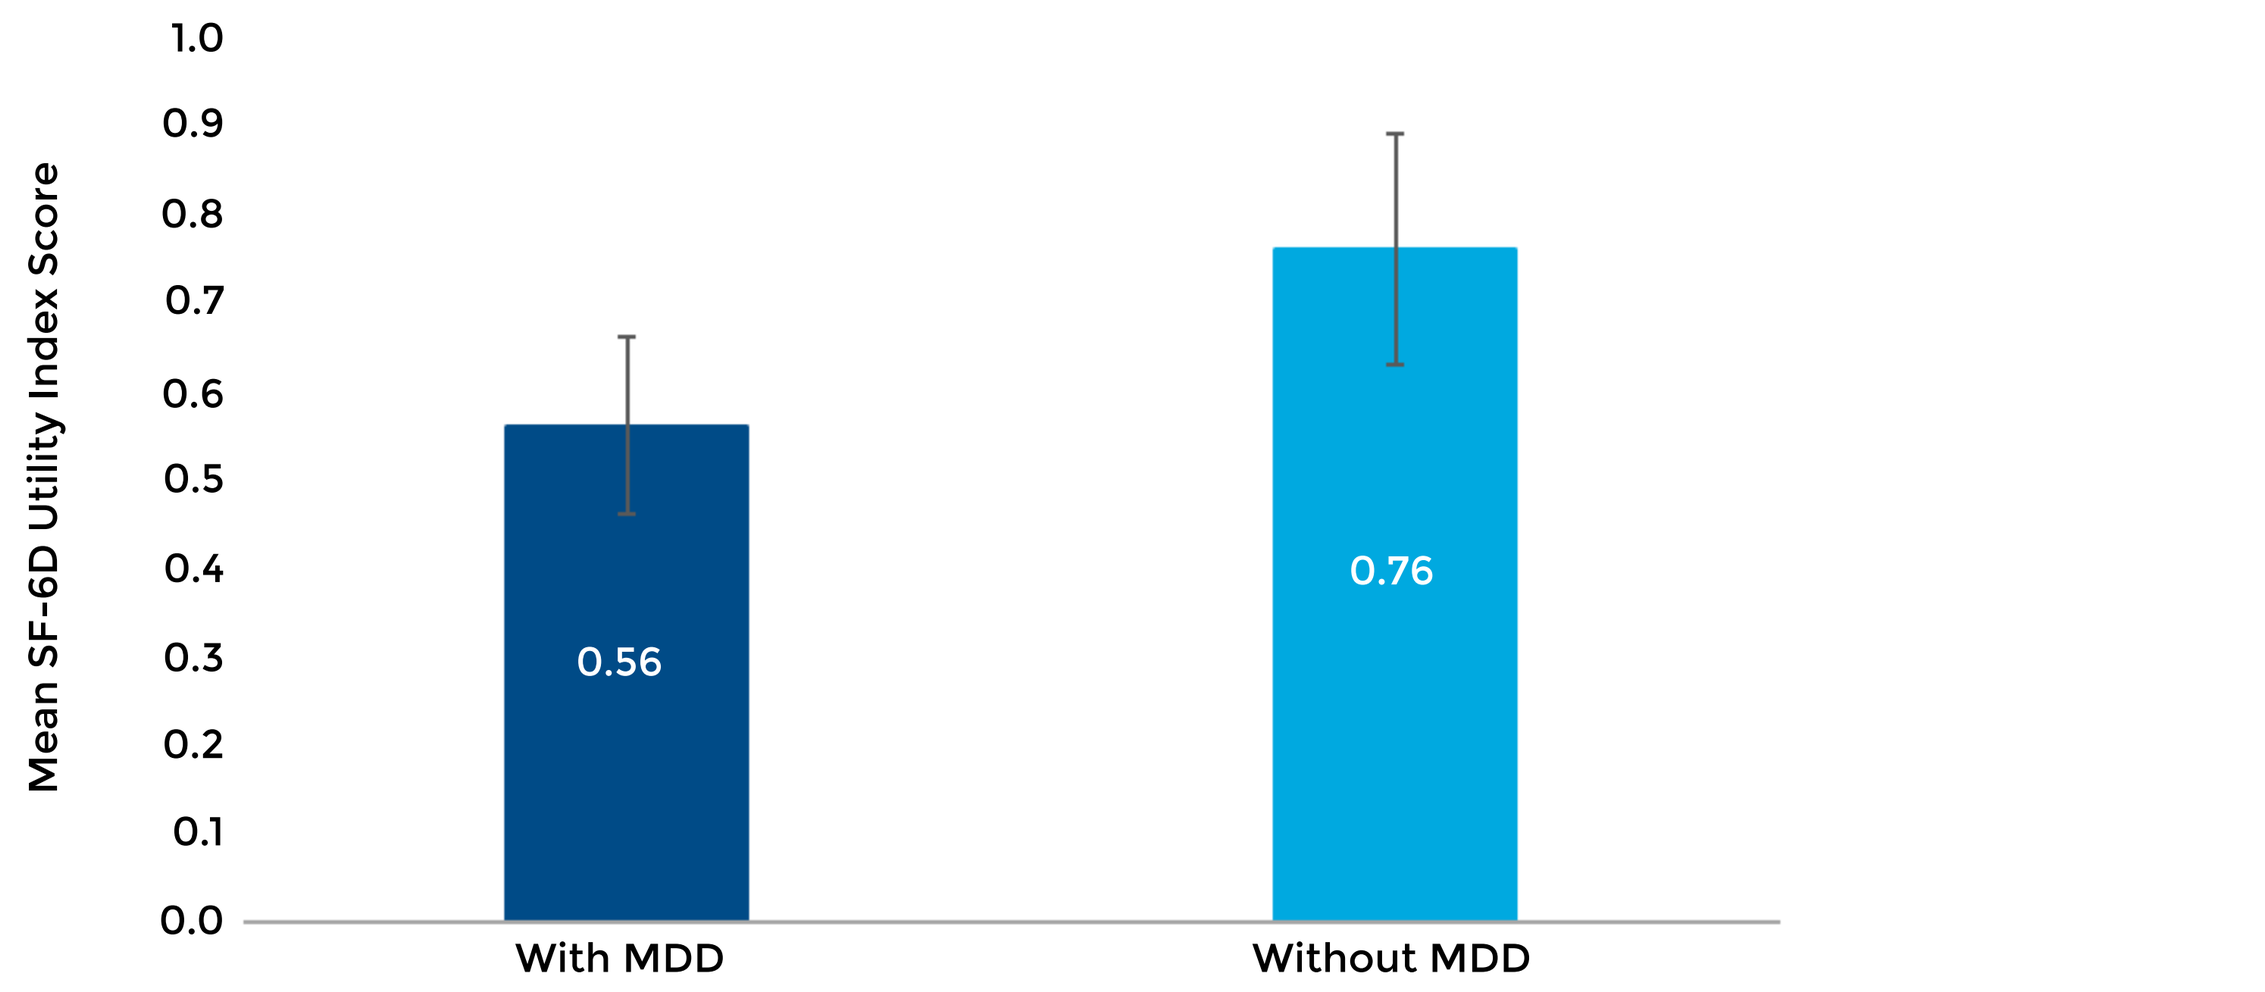
Mean SF-6D Utility Index Score
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0.0
0.56
0.76
With MDD
Without MDD

Figure 3. Social Support was Significantly Lower among Individuals with MDD vs. Individuals without MDD (p <0.001)

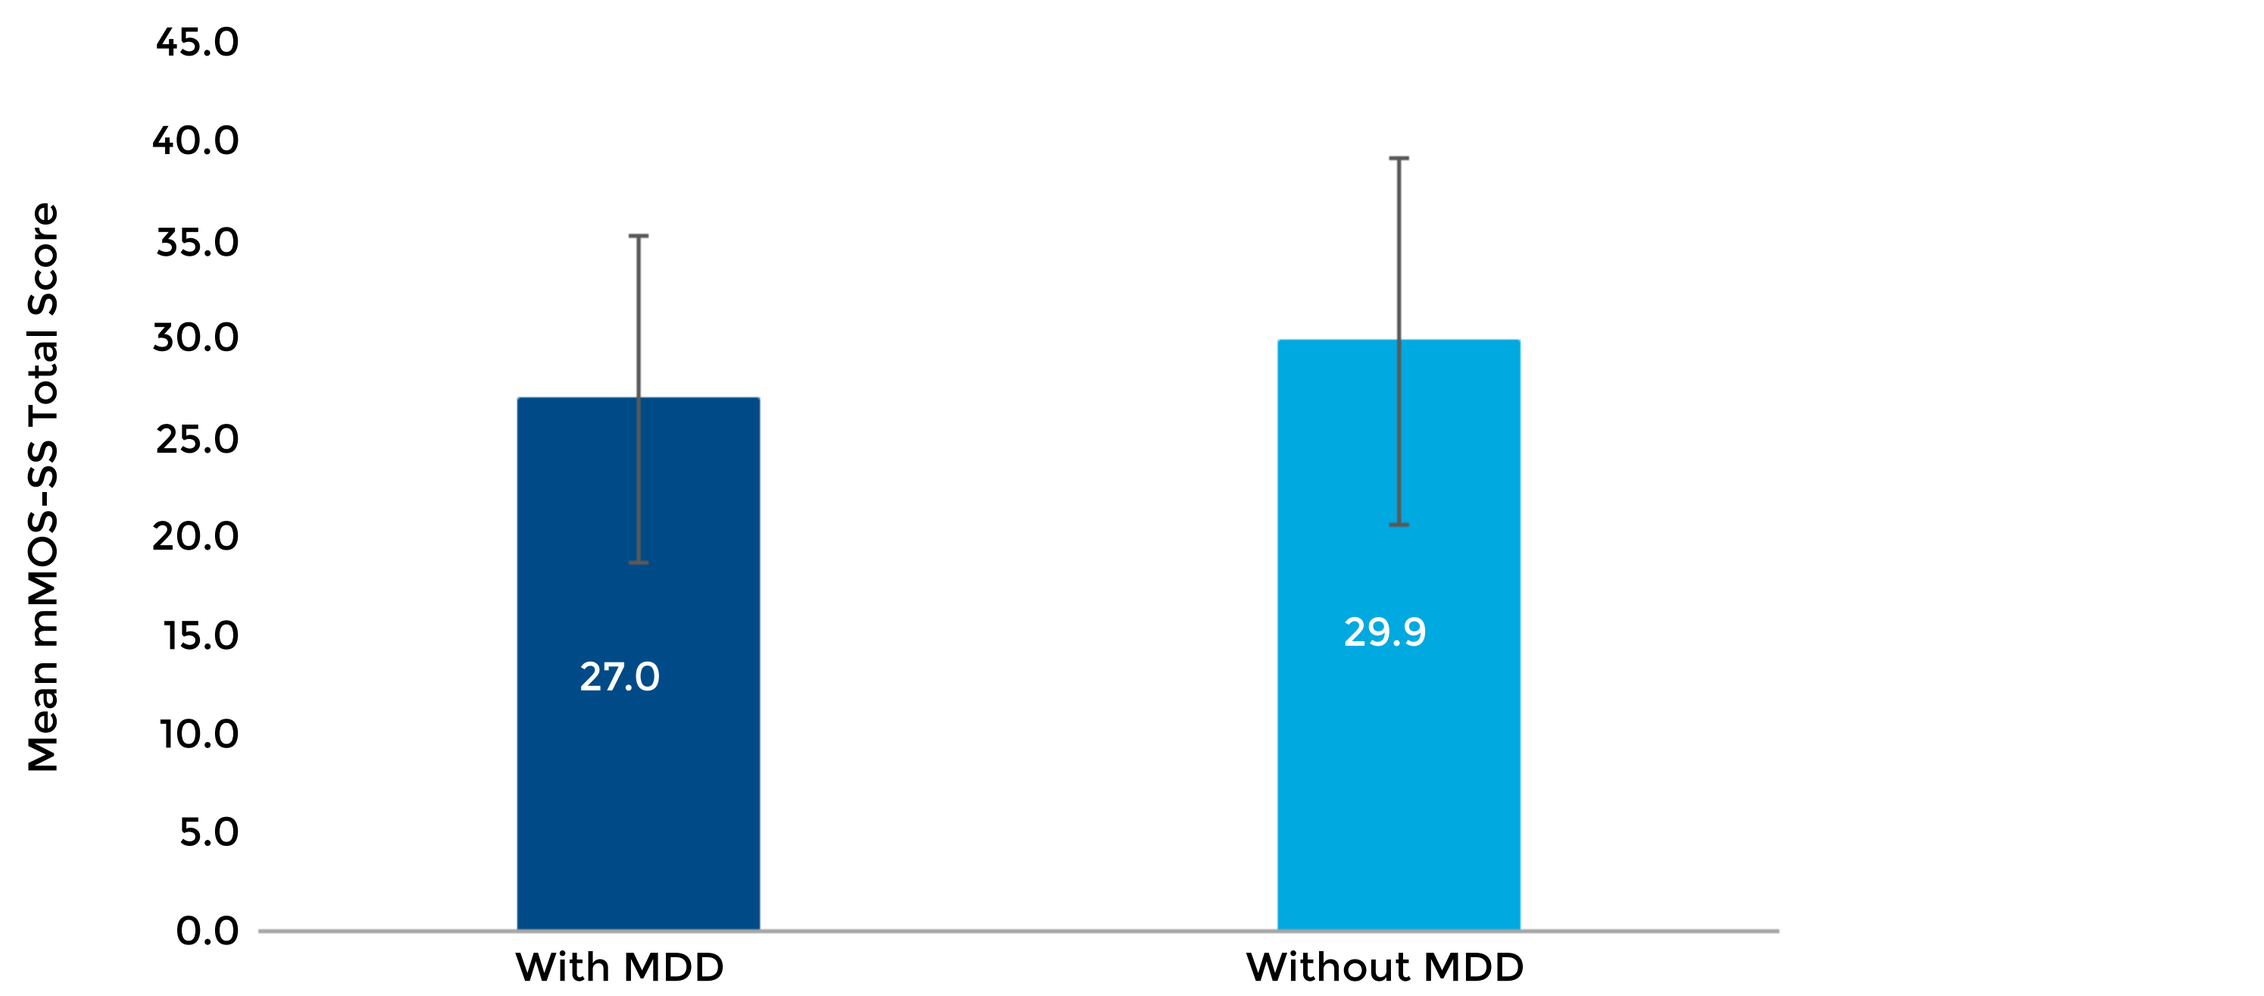
Mean mMOS-SS Total Score
45.0
40.0
35.0
30.0
25.0
20.0
15.0
10.0
5.0
0.0
27.0
29.9
With MDD
Without MDD

Hierarchical (Sequential) Regression Analyses

- A statistically significant three-way interaction between MDD, social support, and race/ethnicity was found (**Table 2**).
- Among individuals with MDD:
 - Social support and QoL were significantly related among individuals who identified as White or 2+ races. However, the relationship between social support and QoL significantly differed among these groups (B= 0.0031, p <0.001) (**Table 3**, **Figure 4**).
 - Among Whites, there was a significant inverse relationship between social support and QoL, such that, as social support increased, QoL decreased (B= -0.0009, p <0.001).
 - Among 2+ races, as social support increased, QoL significantly increased (B= 0.0021, p < 0.01).
- Among individuals without MDD:
 - Social support was associated with a significant positive linear increase in QoL across all race/ethnicity groups (all p-values <0.001) (**Table 3**, **Figure 5**).
 - Compared to Whites, the magnitude of the positive linear association between social support and QoL was significantly lower among Asians (B= -0.0005, p <0.05) and significantly higher among Hispanics (B= 0.0008, p <0.001) (**Table 3**).

Results, continued

Table 2. Results from Hierarchical (Sequential) Regression Model

		R^2	ΔR^2	*p-Value*
Model 1	Covariates	0.1882	–	<0.0001
Model 2	Model 1 + main effects	0.3857	0.1975	<0.0001
Model 3	Model 2 + two-way interactions	0.3929	0.0073	<0.0001
Model 4	Model 3 + three-way interactions	0.3932	0.0003	<0.0001

Note: *p*-values are from the F-tests that were used to compare models.

Table 3. The Estimated Effect of Social Support on QoL and Differences in the Effect of Social Support on QoL, Across Race/Ethnicity Groups and MDD Status

	With MDD		Without MDD	
Estimated Slope (Effect) of Social Support	**Estimate**	**(95% CI)**	**Estimate**	**(95% CI)**
White	-0.0009***	(-0.0012, -0.0007)	0.0024***	(0.0023, 0.0026)
Black/African American	-0.0004	(-0.0009, 0.0002)	0.0023***	(0.0020, 0.0026)
Asian	0.0004	(-0.0005, 0.0012)	0.0019***	(0.0015, 0.0023)
Hispanic	-0.0004	(-0.0008, 0.0000)	0.0032***	(0.0029, 0.0036)
2+ races	0.0021**	(0.0010, 0.0033)	0.0025***	(0.0018, 0.0033)
Estimated Slope (Effect) Difference	**Estimate**	**(95% CI)**	**Estimate**	**(95% CI)**
White	*Ref*	–	*Ref*	–
Black/African American	0.0006	(-0.00005, 0.0012)	-0.0001	(-0.0005, 0.0002)
Asian	0.0013**	(0.0005, 0.0022)	-0.0005*	(-0.0009, -0.0001)
Hispanic	0.0005*	(0.0001, 0.0010)	0.0008***	(0.0005, 0.0012)
2+ races	0.0031***	(0.0019, 0.0042)	0.0001	(-0.0006, 0.0009)

*p <0.05; **p <0.01; ***p <0.001

Note: Estimates were adjusted for gender, marital status, education, census region, smoking status, alcohol use, exercise behaviors, age (in years), and Charlson Comorbidity Index (CCI).

Figure 4. The Estimated Slope (Effect) of Social Support on QoL among Individuals with MDD

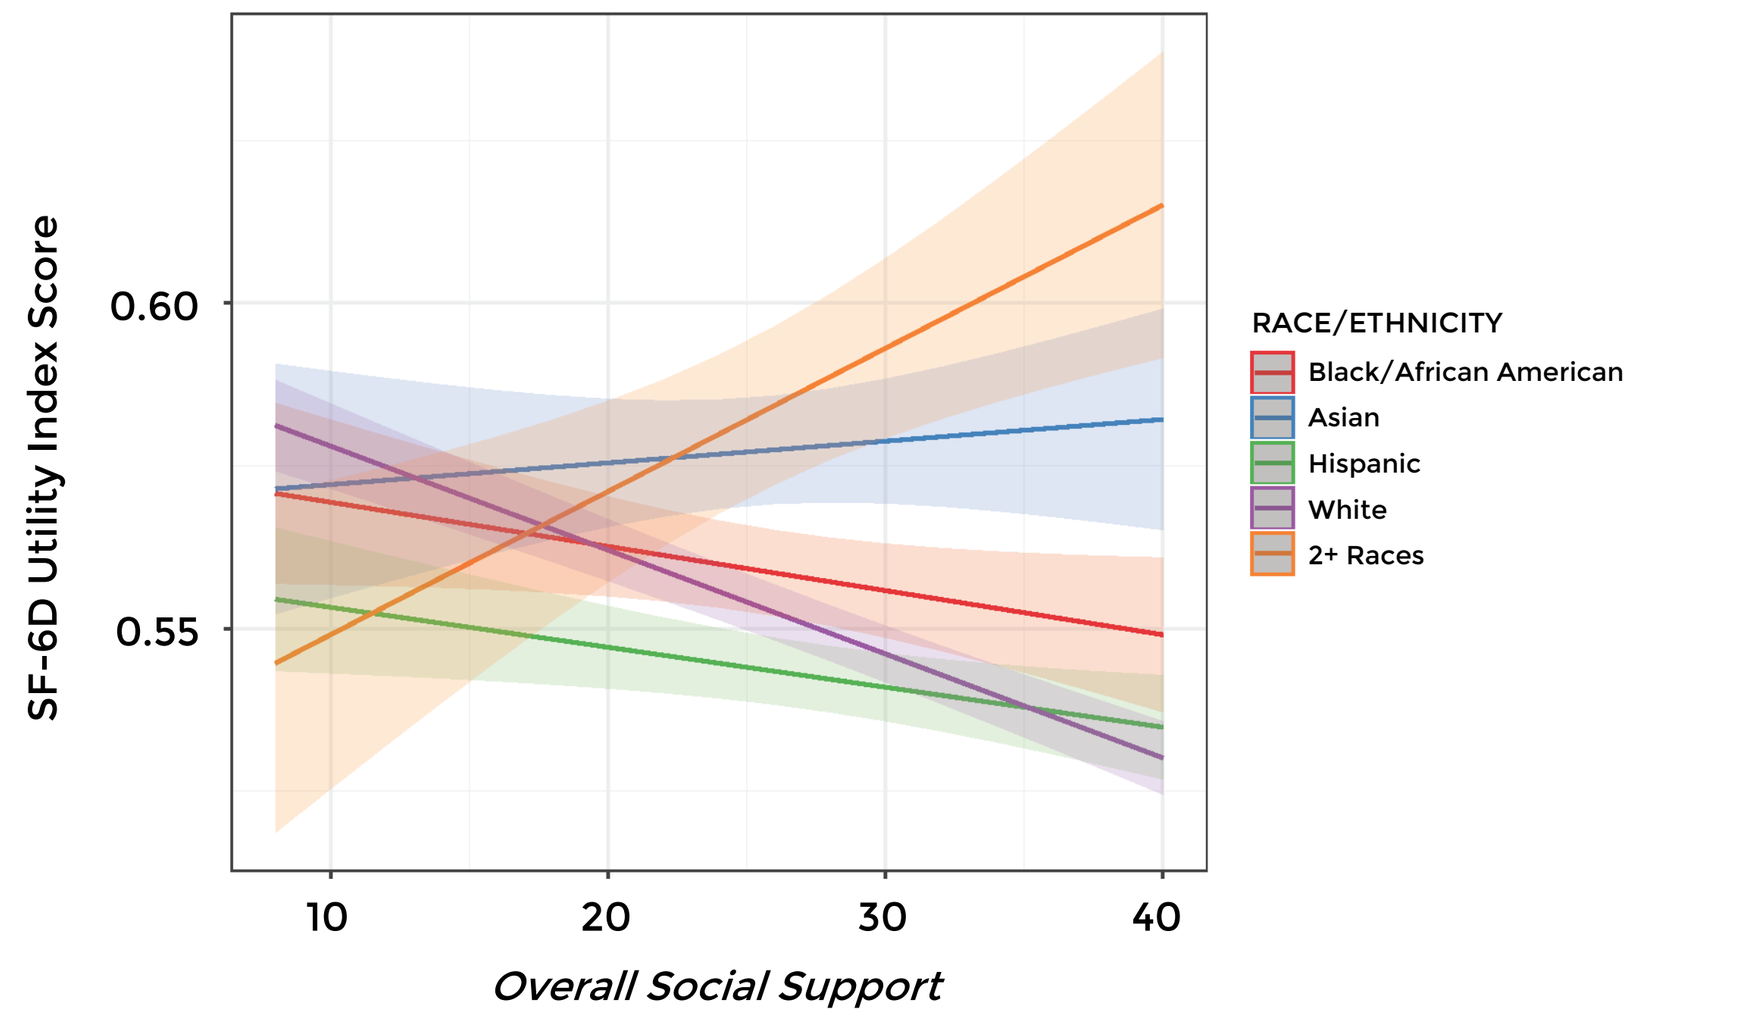
SF-6D Utility Index Score
0.60
0.55
10
20
30
40
Overall Social Support
RACE/ETHNICITY
Black/African American
Asian
Hispanic
White
2+ Races

Figure 5. The Estimated Slope (Effect) of Social Support on QoL among Individuals without MDD

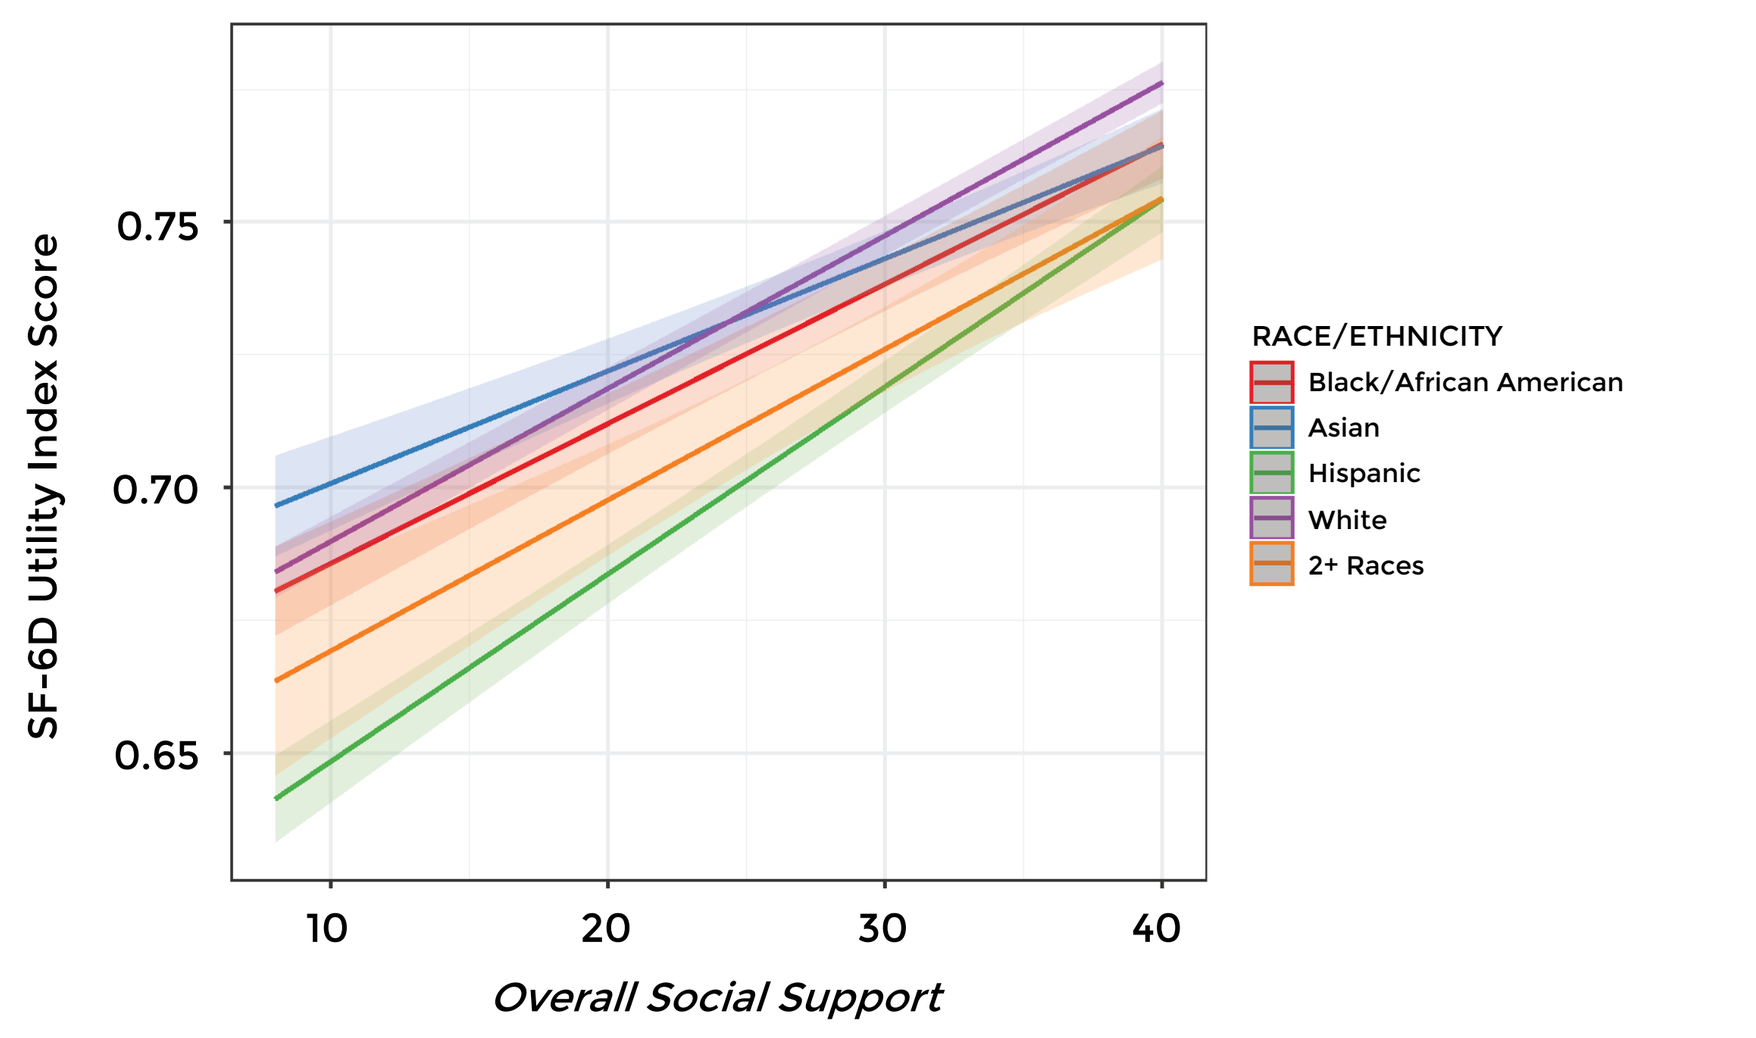
SF-6D Utility Index Score
0.75
0.70
0.65
10
20
30
40
Overall Social Support
RACE/ETHNICITY
Black/African American
Asian
Hispanic
White
2+ Races

Limitations

- The NHWS is cross-sectional; therefore, the temporality of relationships between social support and QoL cannot be determined, nor can changes in such variable be assessed over time.
- While NHWS is representative of the US general adult population, it may not represent the subpopulation with MDD, and the internet-based survey administration may be more likely to overrepresent younger and healthier individuals.

Conclusions

- Using a large, nationally representative study of adults from the US in 2021, we found evidence to suggest social support and race/ethnicity jointly influenced the relationship between MDD status and QoL.
 - With MDD: social support was positively related to QoL among individuals who identified as 2+ races, but inversely related to QoL among individuals who identified as White
 - Without MDD: social support was associated with higher QoL across all race/ethnicity groups
- The relationship between social support and QoL may be bidirectional in the context of serious mental health disorders.
- Understanding the burden of depression may require the consideration of broader contextual factors.

References

1. National Institute of Mental Health. Major Depression. Published January 2022. Accessed March 23, 2022. https://www.nimh.nih.gov/health/statistics/major-depression.
2. Freak-Poli R, Ryan J, Tran T, et al. Social isolation, social support and loneliness as independent concepts, and their relationship with health-related quality of life among older women. https://doi.org/101080/13607863202119400097. Published online 2021. doi:10.1080/13607863.2021.1940097.
3. Richie F, Bonner J, Wittenborn A, Weinstock LM, Zlotnick C, Johnson J. Social Support and Suicidal Ideation (SI) among Prisoners with Major Depressive Disorder (MDD) HHS Public Access. *Arch Suicide Res.* 2021;25(1):107-114. doi:10.1080/13811118.2019.1649773.
4. Bell CN, Thorpe RJ, LaVeist TA. Race/Ethnicity and Hypertension: The Role of Social Support. Published online 2010. doi:10.1038/ajh.2010.28.

Funding

- This study was funded by Cerner Enviza (previously Kantar Health).

Presented at ISPOR 2022, National Harbor, Maryland, USA, May 15-18, 2022

Email: Kathryn.Krupsky@cernerenviza.com
https://www.linkedin.com/in/kkrupsky/