Reasons for Non-Adherence with Antidepressants Using the Medication Adherence Reasons Scale in European Union and United States

Elizabeth Unni,[1] Shaloo Gupta,[2] Nikoletta Sternbach[2]

[1]Touro College of Pharmacy, New York, NY, USA; [2]Cerner Enviza, Malvern, PA, USA

PCR13

Introduction

- Depression is identified by the appearance of sentiments of sadness, desolation, or irritability, along with bodily and cognitive changes continuing for at least 2 weeks, consequentially affecting the individual's competence to function.[1]
- In 2019, 7.2% of European citizens reported having chronic depression and depression costed 118 billion Euros in 2004.[2]
- In 2020, 8.4% of the United States (US) adults had at least one major depressive episode, costing the country $210.5 billion per year.[3]
- Individuals with poor adherence stop taking their medication on an average after 43 days even if the response to the medication is positive and after 15 days if there are adverse effects.[4]
- Understanding the reasons for non-adherence with antidepressants can result in developing targeted interventions to improve the adherence with these medications.
- The Medication Adherence Reasons Scale (MAR-Scale) is a self-report measure that assesses the different aspects of non-adherence, such as reasons and frequency.
- The scale has demonstrated acceptable reliability and validity in 17 disease areas.[5]

Objective

- The study objective was to determine the extent and reasons for non-adherence with antidepressants in the US and 5 European countries (5EU; France, Germany, Italy, Spain, and the UK).

Methods

Data Source and Sample

- Data from the National Health and Wellness Survey (NHWS), a self-administered, internet-based cross-sectional survey of US adults in 2019 (fielded March-July 2019) and 5EU countries in 2020 (fielded December 2019-March 2020) was included.
- NHWS uses a quota sampling framework (sex, age, and race in the US; age and sex in the 5EU) to ensure that it is representative of the demographic composition of the US and 5EU adult population, based on data from the US and International Bureau of the Census.
- The NHWS has been approved by Pearl IRB (Indianapolis, IN, USA).
- NHWS participants who self-reported taking daily prescription medication(s) to treat depression responded to the 19 reasons for non-adherence and one global item of the MAR-Scale. The MAR-Scale measures non-adherence "in the past 7 days," on an 8-point scale ranging from 0 days to 7 days. Frequencies were used to identify the reasons for non-adherence.
- Patient Health Questionnaire (PHQ-9) was used to determine depression severity.[6]

Medication Adherence Reasons Scale (MAR-Scale)

- The scale has 19 items that capture the specific reasons for non-adherence and one global item to measure the extent of self-reported adherence.
 - Respondents were shown the 19 items from the MAR-Scale and were asked to select all the items that were reasons for their non-adherence in the past week. For the items chosen by the respondents, they then selected the number of days, using a 7-point scale (1 day to 7 days) in which that reason was a cause of their non-adherence.

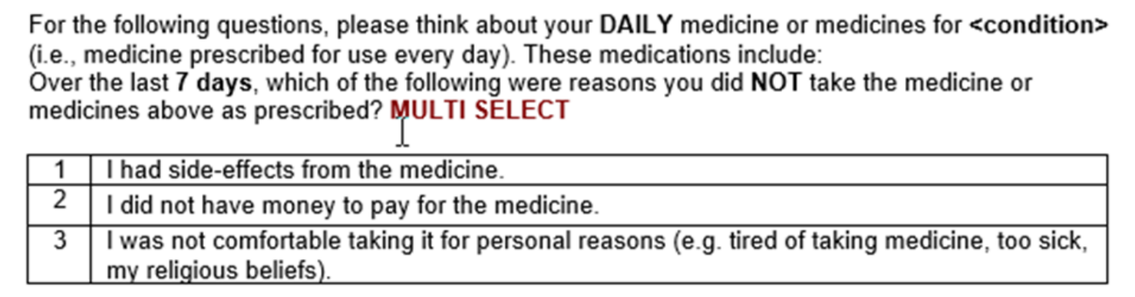
For the following questions, please think about your DAILY medicine or medicines for <condition> (i.e., medicine prescribed for use every day). These medications include:
Over the last 7 days, which of the following were reasons you did NOT take the medicine or medicines above as prescribed? MULTI SELECT

1	I had side-effects from the medicine.
2	I did not have money to pay for the medicine.
3	I was not comfortable taking it for personal reasons (e.g. tired of taking medicine, too sick, my religious beliefs).

- The global item provides an overall estimate of the frequency of medication adherence. Respondents used an 8-point scale (0 days to 7 days) to report the number of days they took the medication as prescribed in the past week.

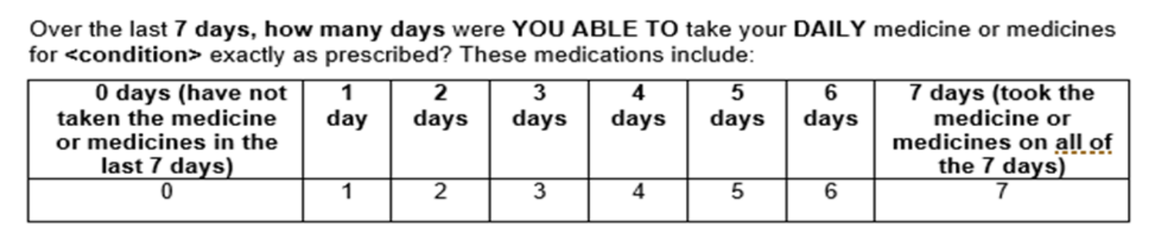
Over the last 7 days, how many days were YOU ABLE TO take your DAILY medicine or medicines for <condition> exactly as prescribed? These medications include:

0 days (have not taken the medicine or medicines in the last 7 days)	1 day	2 days	3 days	4 days	5 days	6 days	7 days (took the medicine or medicines on all of the 7 days)
0	1	2	3	4	5	6	7

Methods, continued

Statistical Analysis

- Descriptive statistics included count and percentages for categorical variables; and means and standard deviations for continuous variables.
- Chi-square tests were used to test differences between groups for categorical variables, and ANOVAs were used to test differences between groups for continuous variables.

Results

Patient Characteristics

- NHWS data had 4,230 patients from the 5EU and 7,506 patients from the US that reported taking a daily antidepressant.
- The 5EU respondents were slightly older than the US respondents, had more annual household income, and had more retirees.
- There were more females and more college graduates among the US respondents.
- While more US respondents were overweight/obese, there were more respondents who smoked and drank among 5EU respondents. Also, more US respondents were currently taking steps to lose weight and had more number of mean days exercising in the past month.

Table 1. Respondent Demographics

Demographics	US 2019 Depression Medication Daily Use (n=7,506)	5EU 2020 Depression Medication Daily Use (n=4,230)	*p-Value*
Age (Mean / SD)	45.86 / 15.87	47.19 / 14.08	<0.001
% Female	72.12	64.18	<0.001
% Annual Household Income <$50K/€50K/£40K	53.25	77.12	<0.001
College Degree or More	38.65	27.87	<0.001
Marital Status			
% Married	38.41	36.69	0.065
% Single, never married	31.35	27.90	<0.001
Retired	16.55	19.13	<0.001

Table 2. Respondent Lifestyle Demographics

Demographics	US 2019 Depression Medication Daily Use (n=7,506)	5EU 2020 Depression Medication Daily Use (n=4,230)	*p-Value*
% Overweight/Obese^	73.43	64.37	<0.001
% Currently Smoke Cigarettes	21.28	37.49	<0.001
% Drink Alcohol	64.18	70.57	<0.001
Mean Days Exercising in the Past Month	5.69	5.14	<0.001
% Currently Taking Steps to Lose Weight	56.21	44.56	<0.001

Note: ^Differing base size – respondents didn't provide weight.

Non-Adherence

- Based on the MAR-Scale, 46.10% of patients reported non-adherence to at least one reason in the 5EU, and 42.86% in the US (**Figure 1**).
- The mean number of days US and 5EU respondents missed taking medication was similar (**Figure 2**).
- In general, individuals who were non-adherent had more severe or moderately severe depression, both in the US (34%) and 5EU (43.5%) (**Figure 3**).
- Across all six countries, patients reported lower non-adherence based on the global question compared to the aggregated score based on the 19 reasons (**Figure 4**).
- Overall, patients in Spain reported the highest non-adherence rate (56.9%, n=240) while patients in the US had the lowest non-adherence rate (42.9%, n=3,217) (**Figure 4**).

Results, continued

Top Reasons for Non-Adherence

- The most common reason for non-adherence in the 5EU was concern about long-term effects (23.92%) and possible side effects from the medication (20.40%).
- In the US, though simple forgetfulness (20.86%) was the major reported reason for non-adherence, patients reported the lowest mean number of days (2.15 days/week) missing medication for that reason.
- Non-adherence lasted longer due to lack of beliefs in needing medication anymore, 4.06 days/week in the US and 3.32 days/week in the 5EU.
- For 11 of the 19 reasons, respondents missed their medication for more than 3 days a week in the US. There were only 6 reasons for which respondents missed their medication for more than 3 days a week in the 5EU.
- Cost was an issue for 8% of US respondents and 7% of 5EU respondents to miss their medication for more than 3 days/week.
- Though only a small percentage of the respondents missed their medication due to reasons such as not knowing how to take the medication, difficulty swallowing the medication, or opening the container, these reasons caused them to miss their medication for more than 3 days/week.

Figure 1. Non-Adherence – Aggregated Score from 19 Reasons

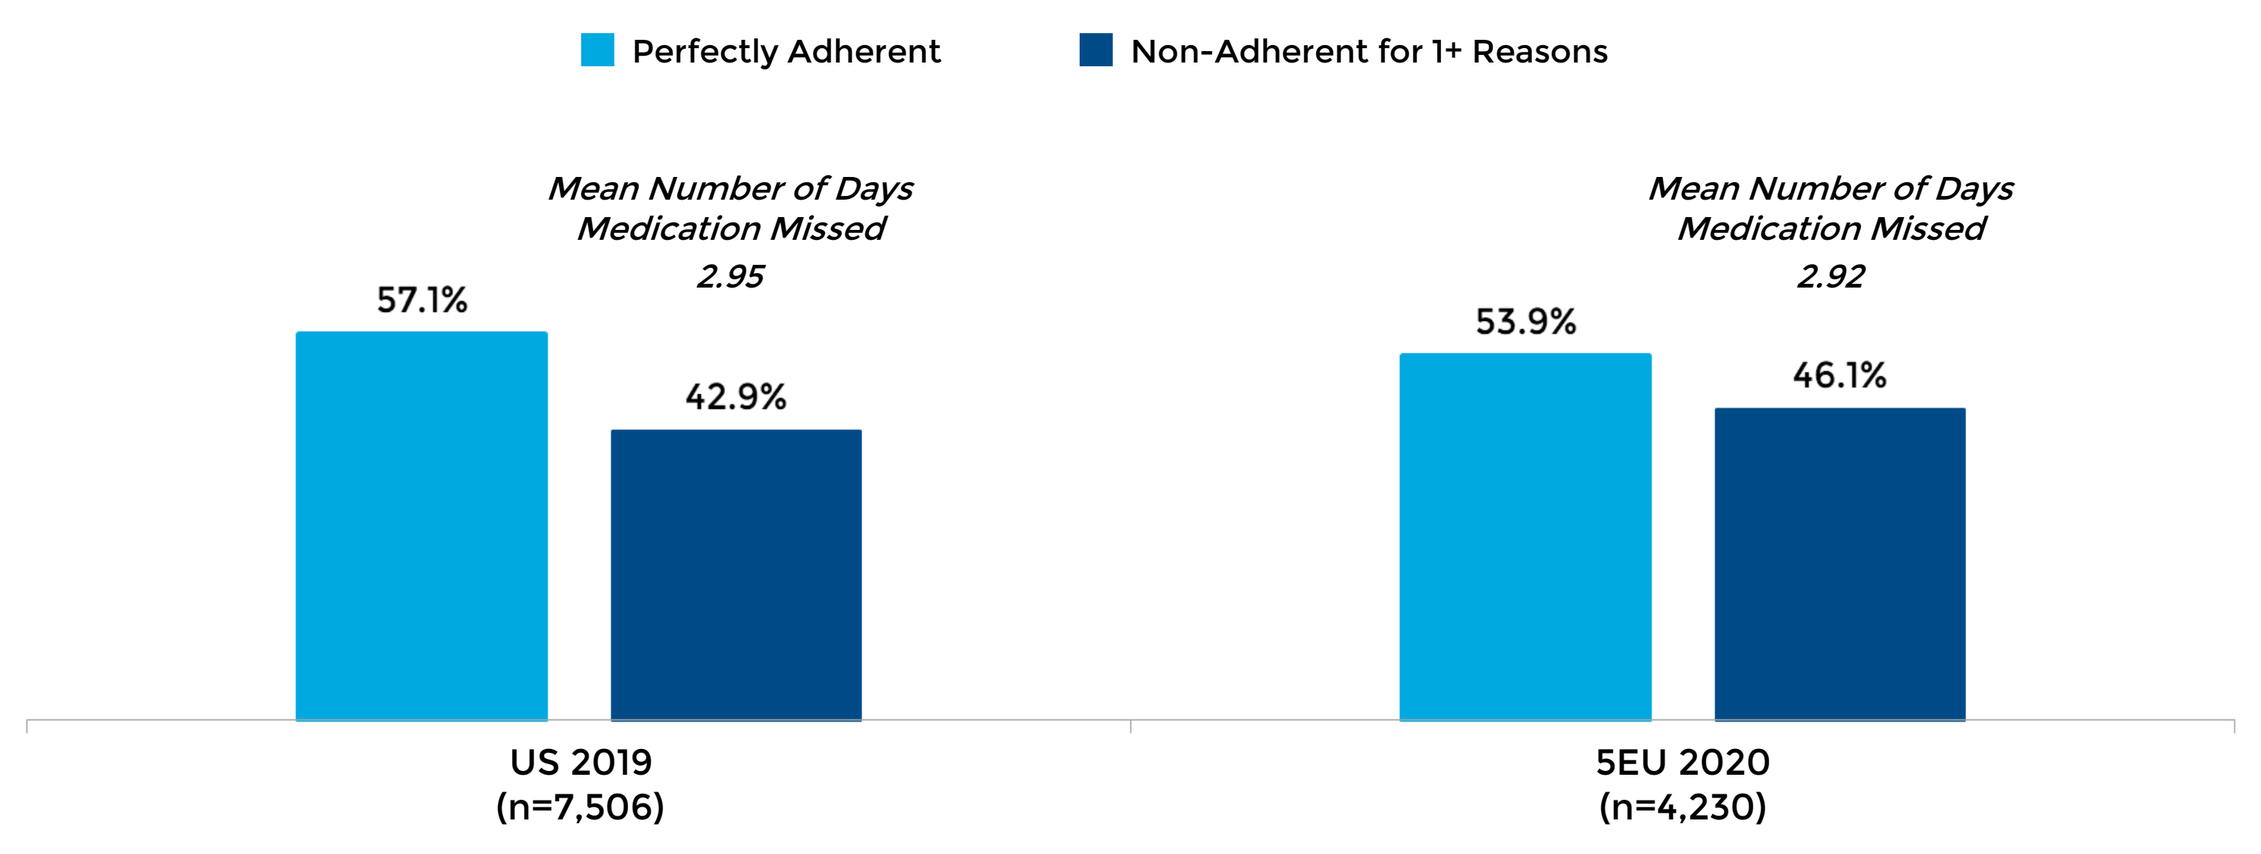

Perfectly Adherent
Non-Adherent for 1+ Reasons
Mean Number of Days Medication Missed 2.95
57.1%
42.9%
US 2019 (n=7,506)
Mean Number of Days Medication Missed 2.92
53.9%
46.1%
5EU 2020 (n=4,230)

Figure 2. Top Reasons for Medication Non-Adherence in Depression

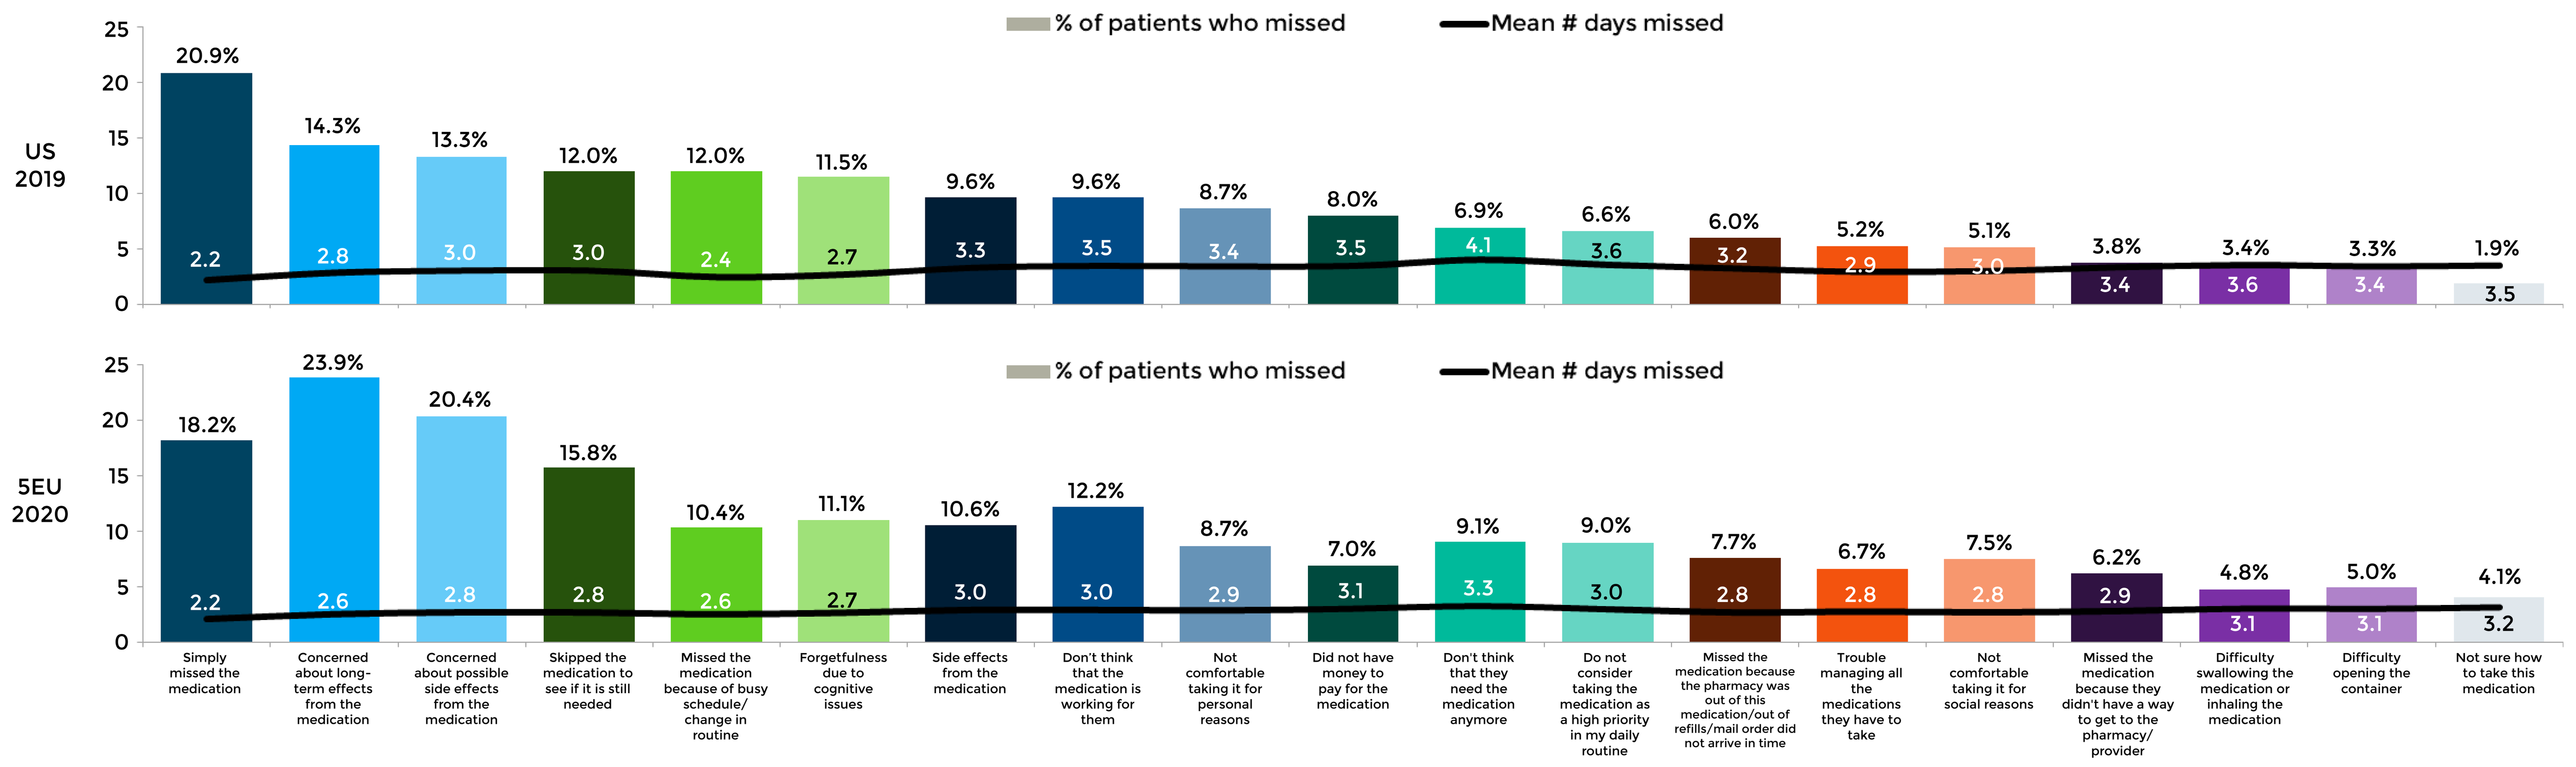

% of patients who missed
Mean # days missed
US 2019
20.9% 2.2; 14.3% 2.8; 13.3% 3.0; 12.0% 3.0; 12.0% 2.4; 11.5% 2.7; 9.6% 3.3; 9.6% 3.5; 8.7% 3.4; 8.0% 3.5; 6.9% 4.1; 6.6% 3.6; 6.0% 3.2; 5.2% 2.9; 5.1% 3.0; 3.8% 3.4; 3.4% 3.6; 3.3% 3.4; 1.9% 3.5
5EU 2020
18.2% 2.2; 23.9% 2.6; 20.4% 2.8; 15.8% 2.8; 10.4% 2.6; 11.1% 2.7; 10.6% 3.0; 12.2% 3.0; 8.7% 2.9; 7.0% 3.1; 9.1% 3.3; 9.0% 3.0; 7.7% 2.8; 6.7% 2.8; 7.5% 2.8; 6.2% 2.9; 4.8% 3.1; 5.0% 3.1; 4.1% 3.2
Simply missed the medication
Concerned about long-term effects from the medication
Concerned about possible side effects from the medication
Skipped the medication to see if it is still needed
Missed the medication because of busy schedule/change in routine
Forgetfulness due to cognitive issues
Side effects from the medication
Don't think that the medication is working for them
Not comfortable taking it for personal reasons
Did not have money to pay for the medication
Don't think that they need the medication anymore
Do not consider taking the medication as a high priority in my daily routine
Missed the medication because the pharmacy was out of this medication/out of refills/mail order did not arrive in time
Trouble managing all the medications they have to take
Not comfortable taking it for social reasons
Missed the medication because they didn't have a way to get to the pharmacy/provider
Difficulty swallowing the medication or inhaling the medication
Difficulty opening the container
Not sure how to take this medication

Figure 3. Percentage of Respondents at Different Depression Severity Levels via PHQ-9

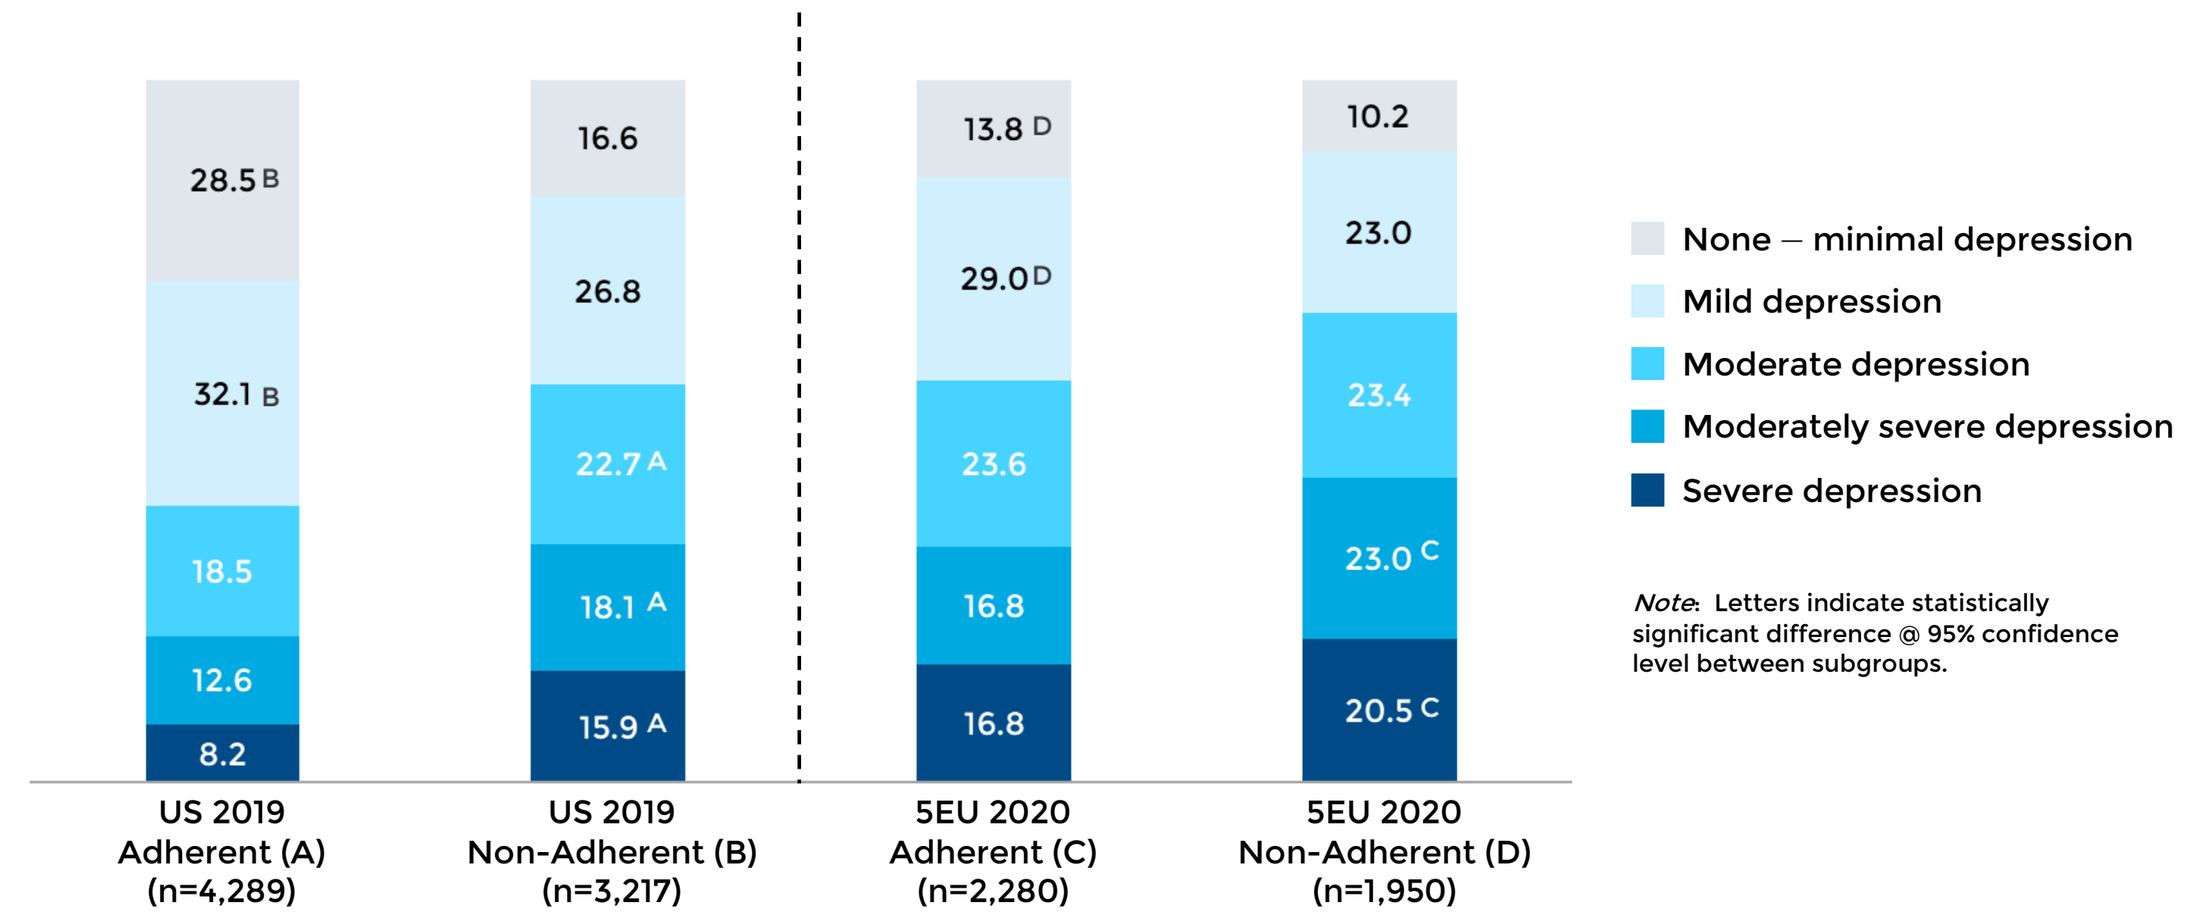

US 2019 Adherent (A) (n=4,289): 28.5 B; 32.1 B; 18.5; 12.6; 8.2
US 2019 Non-Adherent (B) (n=3,217): 16.6; 26.8; 22.7 A; 18.1 A; 15.9 A
5EU 2020 Adherent (C) (n=2,280): 13.8 D; 29.0 D; 23.6; 16.8; 16.8
5EU 2020 Non-Adherent (D) (n=1,950): 10.2; 23.0; 23.4; 23.0 C; 20.5 C
None – minimal depression
Mild depression
Moderate depression
Moderately severe depression
Severe depression
Note: Letters indicate statistically significant difference @ 95% confidence level between subgroups.

Figure 4. Non-Adherent Rate Based on MAR-Scale

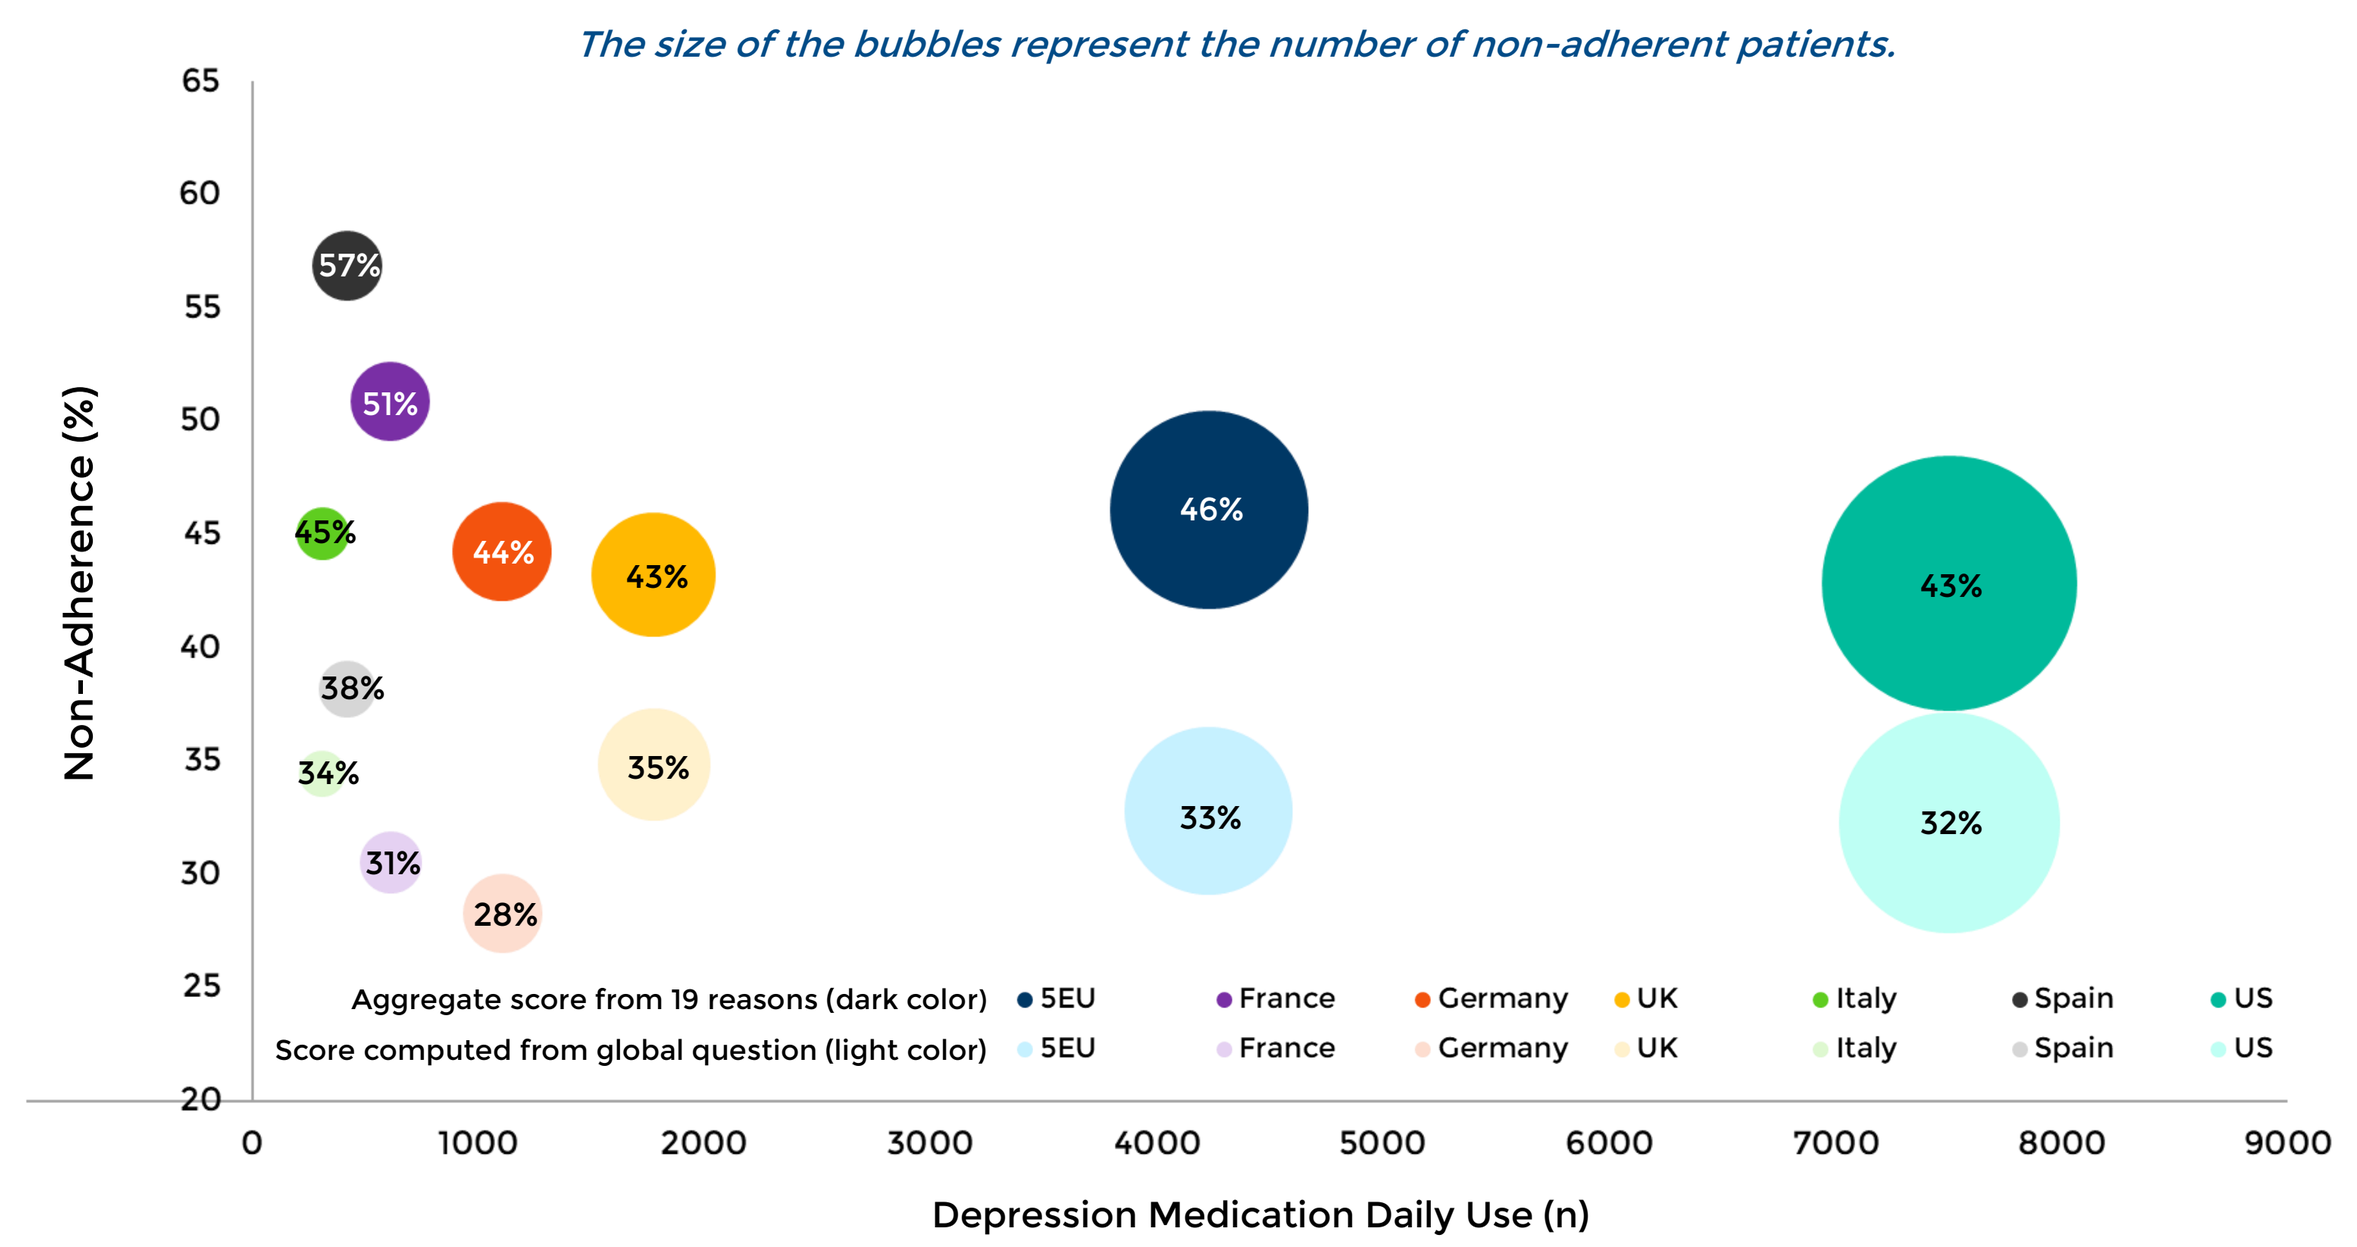

The size of the bubbles represent the number of non-adherent patients.
Non-Adherence (%)
57% 51% 45% 44% 43% 46% 43% 38% 35% 34% 33% 32% 31% 28%
Aggregate score from 19 reasons (dark color): 5EU, France, Germany, UK, Italy, Spain, US
Score computed from global question (light color): 5EU, France, Germany, UK, Italy, Spain, US
Depression Medication Daily Use (n)

Discussion

- The study results of non-adherence rates at 43% in the US and 46% in the 5EU are comparable to results from other studies.[7]
- The reason for non-adherence are also comparable between the US and the 5EU. Approximately, one in five are missing medication due to simple forgetfulness. Concern belief about the medication and lack of necessity beliefs with the medication are major reasons for non-adherence.
- Individuals often overestimate their level of adherence as can be seen from the difference between the adherence score computed from the global item and the aggregate of 19 reasons. Clinicians need to ask specific questions about adherence to develop interventions.
- Cost is still an issue for missing the medication for half the time.
- Non-adherence is correlated with the severity of depression. More non-adherents are also severely depressed.

Limitations

- Analyses are based on self-report data of adherence measures. Recall bias may introduce error, in addition to overestimation and/or underestimation of adherence.
- Cross-sectional studies account for only one point in time and do not capture adherence behavior over time. Replicating these results with a longitudinal study would be beneficial.

Conclusions

- With antidepressants, further interventions are needed to educate the patients about the need for the medications and to lessen their concerns about long-term effects and possible side effects from the medications.
- Patients with depression may also benefit from reminder interventions.
- Efforts should also be taken to teach individuals how to take medications and reduce logistic issues such as opening containers or difficulty swallowing.

References

1. Villarroel MA, Terlizzi EP. Symptoms of Depression among Adults: United States, 2019. *NCHS Data Brief*. 2020;(379):1-8.
2. Sobocki P, Jönsson B, Angst J, Rehnberg C. Cost of depression in Europe. *J Ment Health Policy Econ*. 2006 Jun;9(2):87-98. PMID: 17007486. https://ec.europa.eu/eurostat/web/products-eurostat-news/-/edn-20210910-1.
3. Greenberg, PE, Fournier AA, Sisitsky T, Pike CT & Kessler RC. (2015). The economic burden of adults with major depressive disorder in the United States (2005 and 2010). *Journal of Clinical Psychiatry*, 76, 155-162. doi: 10.4088/JCP.14m09298. https://www.nimh.nih.gov/health/statistics/major-depression.
4. Linden M, Gothe H, Dittmann RW, et al. Early termination of antidepressant drug treatment. *J. Clin. Psychopharm* 20(5), 523-530 (2000).
5. Unni EJ, Sternbach N, Goren A. Using the Medication Adherence Reasons Scale (MAR-Scale) to identify the reasons for non-adherence across multiple disease conditions. *Patient Prefer Adherence*. 2019 Jun 28;13:993-1004.
6. Kroenke K, Spitzer RL, Williams JBW. The PHQ-9: Validity of a brief depression severity measure. *J Gen Intern Med*. 2001;16:606-613.
7. Vazquez DMJM. (2017). Adherence to Antidepressants. A Review of the Literature. *Mathews J Psychiatry Ment Health*. 2(1):009.

Presented at ISPOR 2022, National Harbor, Maryland, USA, May 15-18, 2022

Contact Elizabeth Unni for the use of MAR-Scale (elizabeth.unni@touro.edu)