Presentation Number 114701

Healthcare resource utilization in the treatment of transfusion-dependent ß-thalassemia

Russell L. Knoth,[1] Kacper Perkowski,[2] Halley Costantino,[2] Brian Inyart,[2] Lauren Ashka,[2] Kelly Clapp,[2] Shaloo Gupta[2]

[1]Bristol Myers Squibb, Princeton, NJ; [2]Cerner Enviza, Malvern, PA; USA

Introduction

- ß-thalassemia is a genetic blood disorder characterized by inadequate hemoglobin production
- Many patients develop chronic anemia that requires ongoing red blood cell transfusions (RBCTs)[1]
- Transfusion-dependent ß-thalassemia (TDT) has been shown to negatively impact health-related quality of life and psychosocial outcomes[2-5]
- However, the impact of TDT on healthcare resource utilization (HCRU) and costs has not been well characterized

Objective

- To examine HCRU and costs among patients with TDT in the United States

Methods

- Cooley's Anemia Foundation identified and recruited adults (≥ 18 years of age) with ß-thalassemia in May 2021
 - Eligible patients had a self-reported physician diagnosis of ß-thalassemia and had received ≥ 1 RBCT in the past 6 months
- Patients completed a questionnaire reporting their demographics, disease characteristics, treatment, HCRU, and direct and indirect costs
 - Productivity outcomes were assessed using the Work Productivity and Activity Impairment (WPAI) instrument[6]
- All patients provided informed consent; the study materials were reviewed and approved by Pearl Institutional Review Board (Indianapolis, IN, USA)

Statistical analysis

- All findings were analyzed using descriptive statistics

Results

Patient characteristics

- A total of 100 patients completed the survey; mean age was 35.99 years (standard deviation [SD], 10.36) and 65% were female (**Table 1**)
- 98% of patients had health insurance; 57.1% were commercially insured
- Patients had been diagnosed with ß-thalassemia for a mean of 34.40 years (SD, 10.33)
- Overall, patients experienced a mean of 9.62 RBCTs (SD, 4.32) in the previous 6 months
- Nearly all patients (97%) had ≥ 1 RBCT per month; most (66%) had > 1 RBCT per month
- Patients typically also received iron chelation therapy (94%) and surgery (47%)

Table 1. Demographic and clinical characteristics

Characteristic	Patients (N = 100)
Age, mean (SD), years	35.99 (10.36)
Female, n	65
Race/ethnicity, n	
Asian or Pacific Islander	42
White	34
Hispanic	14
Mixed race/other	9
Declined to answer	1
Employed, n	63
Marital status, n	
Married/living with partner	46
Single/never married	49
Separated/divorced/widowed	5
Household income, n	
USD ≤ 99,999	54
USD ≥ 100,000	33
Declined to answer	13
Education, n	
College degree or higher	69
Health insurance, n (%)[a]	n = 98
Managed care/HMO/PPO	56 (57.1)
Medicare	23 (23.5)
Medicaid	23 (23.5)
High deductible health plan with HSA	5 (5.1)
Other	11 (11.2)
Top 5 comorbidities, n	
Anemia	33
Osteoporosis	33
Anxiety	18
Migraines/headaches	11
Depression	10
Charlson comorbidity index, mean (SD)	0.58 (1.23)
Charlson comorbidity index, n	
0	76
1	7
≥ 2	17
Time since diagnosis, mean (SD), years	34.40 (10.33)
Number of RBCTs in the past 6 months, mean (SD)	9.62 (4.32)
Frequency of RBCT, n	
> 1 per month	66
Monthly	31
Every other month	3

[a]Patients could select more than one type of health insurance.
HMO, health maintenance organization; HSA, health savings account; PPO, preferred provider organization; USD, United States dollar.

HCRU

- Nearly all patients had a ß-thalassemia-related physician office visit (92%) and laboratory/blood tests (83%), excluding for RBCTs, in the previous 6 months (**Table 2**)
- 7% of patients had ß-thalassemia-related emergency department (ED) visits and 3% had hospitalizations
- 39% of patients reported a high or extremely high burden of insurance-related issues

Table 2. Patient-reported HCRU

Healthcare encounter	Patients[a]
ß-thalassemia-related office visit excluding RBCT/previous 6 months, n	92
Length of office visit, n (%), minutes[b]	n = 92
< 15	10 (10.9)
15-30	14 (15.2)
31-45	18 (19.6)
46-60	25 (27.2)
61-90	14 (15.2)
> 90	11 (12.0)
ß-thalassemia-related laboratory/blood test excluding RBCT/previous 6 months, n	83
Length of ß-thalassemia-related laboratory/blood test visits, n (%), minutes[b]	n = 83
< 10	14 (16.90)
11-20	20 (24.10)
21-30	27 (32.50)
31-60	16 (19.30)
>60	6 (7.20)
ß-thalassemia-related ED visit/previous 6 months, n	7
Length of ß-thalassemia-related ED visits, n (%), hours[b]	n = 7
< 1	1 (14.30)
1-2	0 (0.00)
>2-4	1 (14.30)
>4-6	3 (42.90)
6+	2 (28.60)
ß-thalassemia-related hospitalization/previous 6 months, n	3
Length of ß-thalassemia-related hospitalization, n (%), days[b]	n = 3
1-2	2 (66.70)
3-4	1 (33.30)
Insurance-related burden, n[b]	n = 100
Extremely high burden	11
High burden	28
Moderate burden	15
Some burden	22
No burden	24

[a]Percentages may not total 100% due to rounding.
[b]For those with ≥ 1 corresponding healthcare encounter or with health insurance.

Direct costs

- Self-reported expenses were most often related to transportation and medications (**Figure 1**)
- Patients reported substantial direct costs both for medical encounters and expenses outside of care (**Table 3**)
 - Mean 6-month out-of-pocket costs were USD 555.23 (SD, 1981.87) for office visits and USD 450.70 (SD, 1531.11) for laboratory/blood tests both not including RBCT-related visits, for those with ≥ 1 corresponding healthcare encounter

Indirect costs

- Of the 63 patients who were employed, 82.5% reported that their employment decisions were impacted by their ß-thalassemia
 - How choice of employment was affected is described in **Figure 2**

Figure 1. Percentage of patients who incurred expenses outside of TDT care

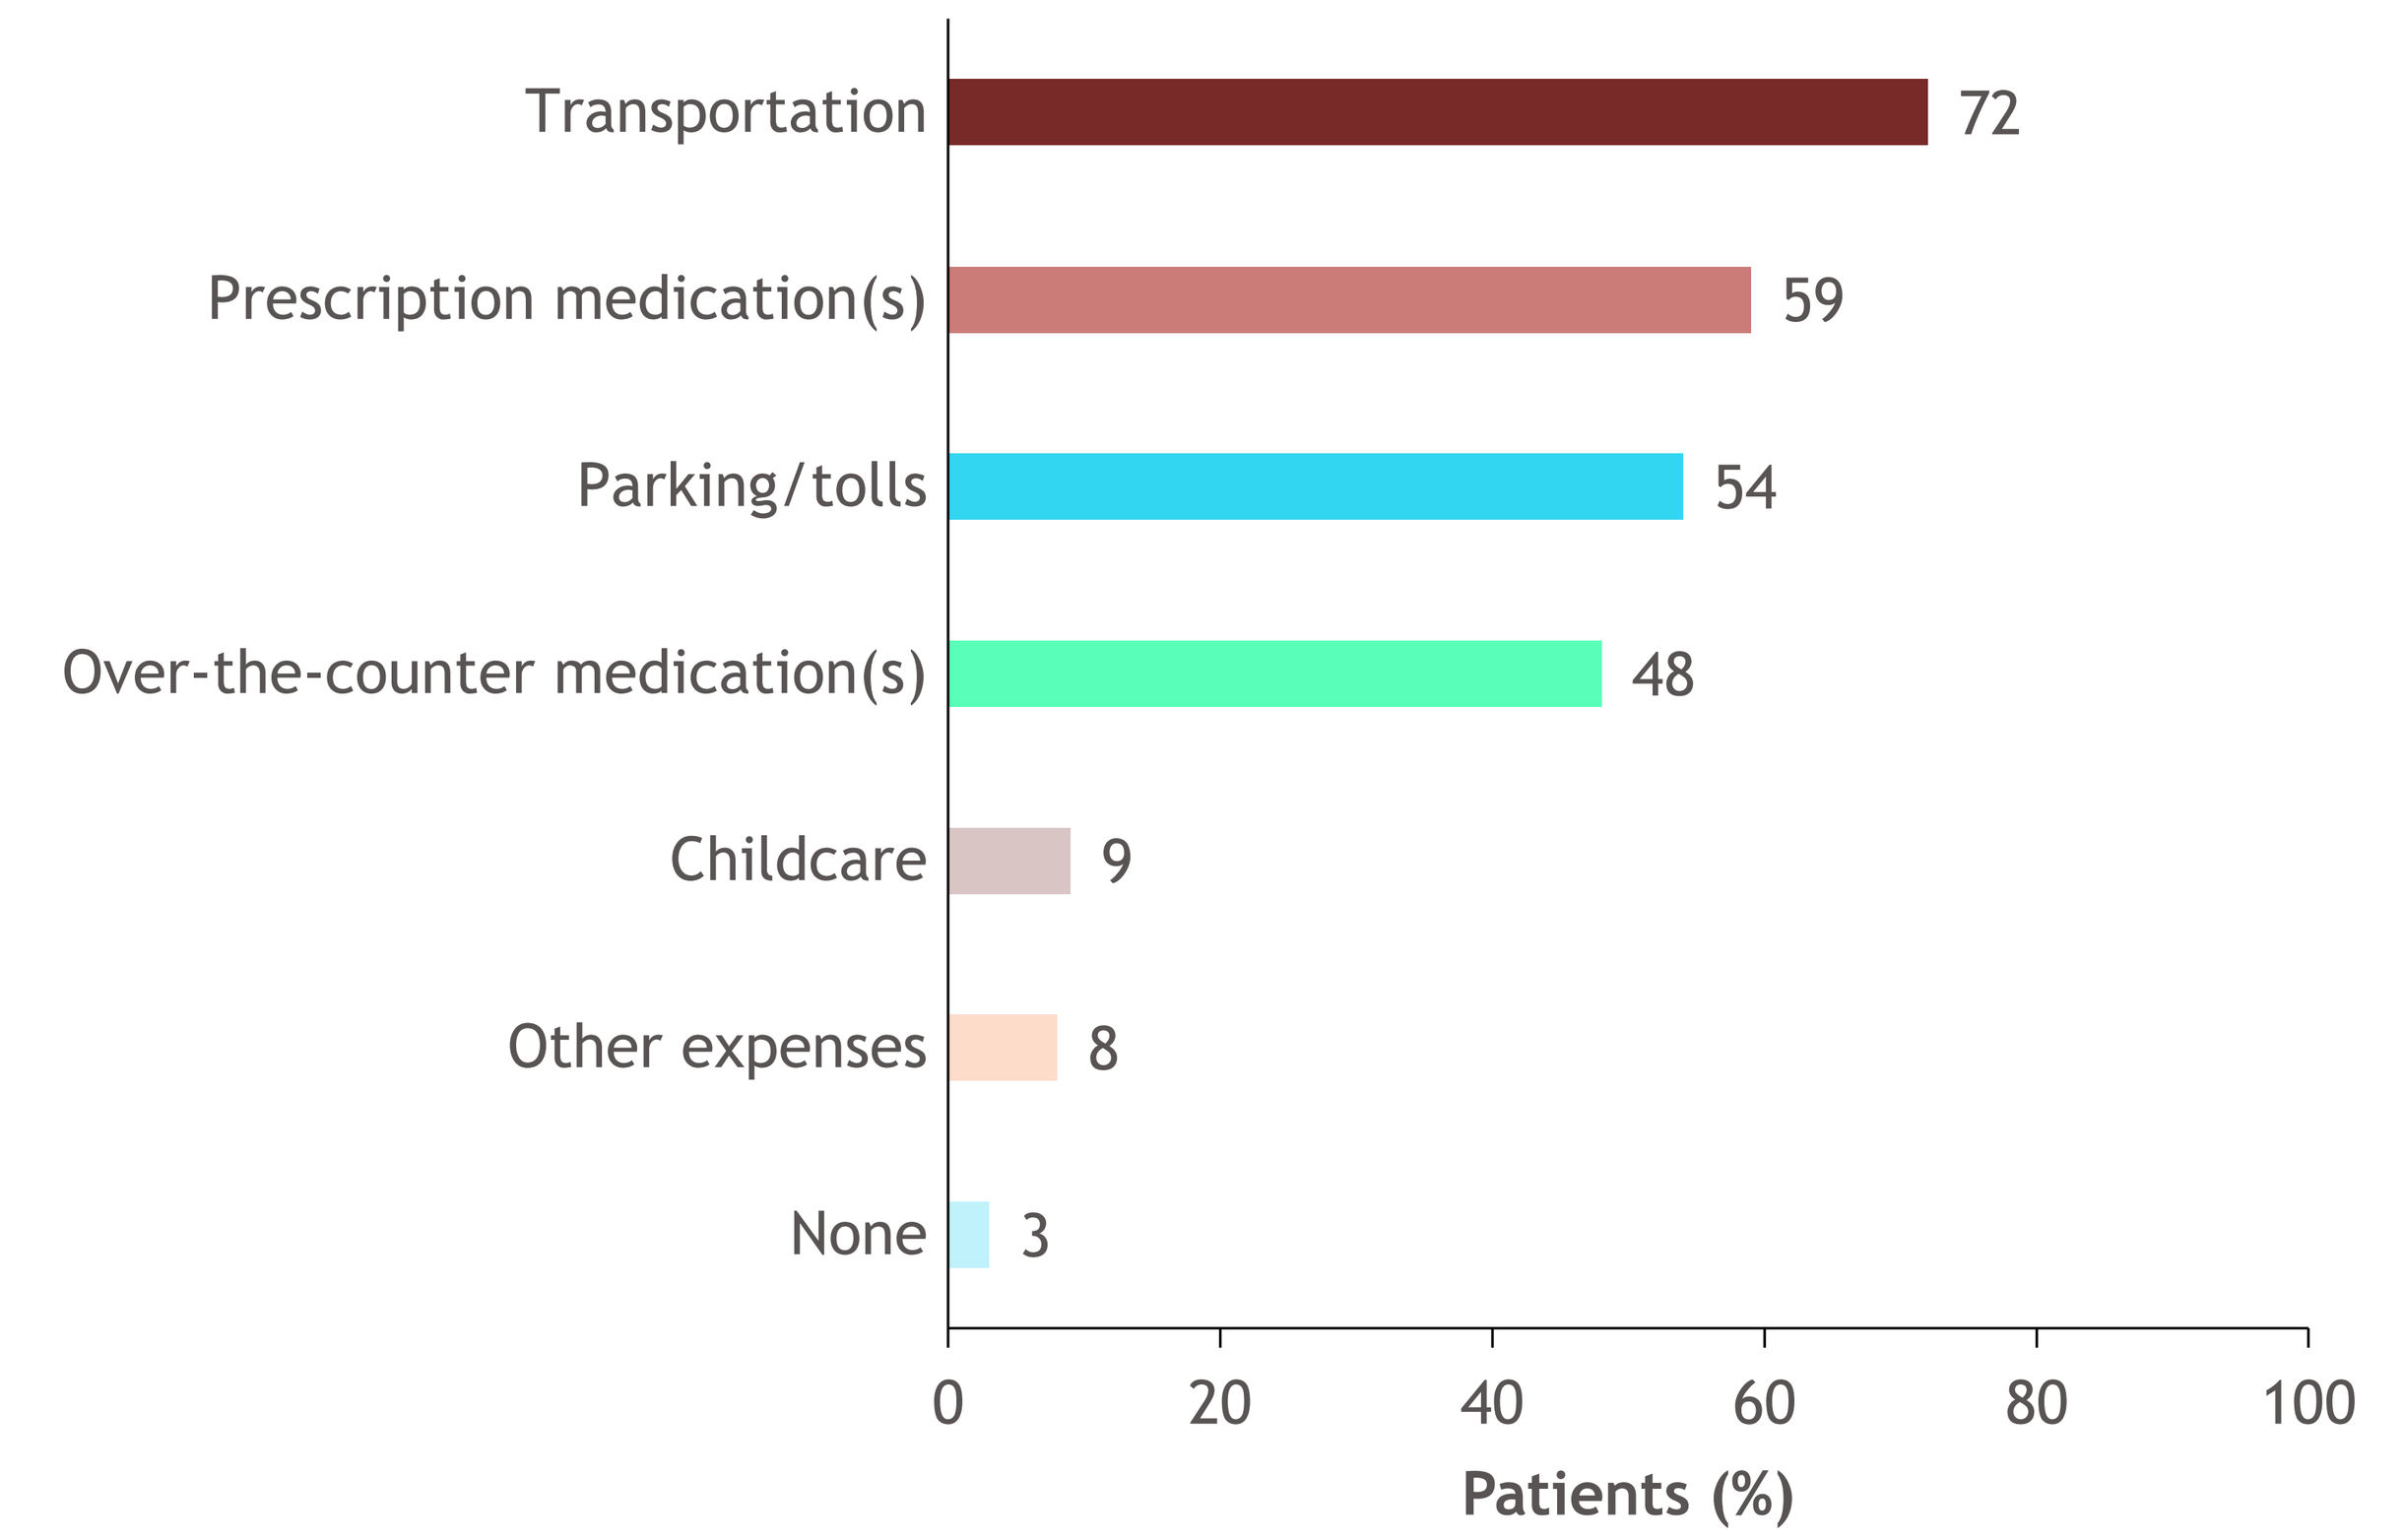
Transportation
72
Prescription medication(s)
59
Parking/tolls
54
Over-the-counter medication(s)
48
Childcare
9
Other expenses
8
None
3
0
20
40
60
80
100
Patients (%)

Table 3. Patient-reported out-of-pocket direct costs[a]

Healthcare encounter costs	Patients	1 visit cost (SD), USD	6 months cost (SD), USD
ß-thalassemia-related office visits excluding RBCT	92	97.13 (272.37)	555.23 (1981.87)
ß-thalassemia-related laboratory/ blood tests excluding RBCT	83	68.33 (218.20)	450.70 (1531.11)
ß-thalassemia-related ED visit/ previous 6 months	7	143.00 (153.76)	143.00 (153.76)
ß-thalassemia-related hospitalization/ previous 6 months	3	41.67 (52.04)	41.67 (52.04)
Costs outside of TDT care	**Patients**	**1 month cost (SD), USD**	**6 months cost (SD), USD**
Transportation	72	29.81 (29.61)	178.83 (177.64)
Prescription medication(s)	59	131.93 (416.84)	791.59 (2501.01)
Parking/tolls	54	18.50 (21.92)	111.00 (131.50)
Over-the-counter medication(s)	48	26.25 (32.01)	157.50 (192.06)
Childcare	9	296.67 (463.11)	1780.00 (2778.69)
Other expenses	8	147.50 (264.09)	885.00 (1584.53)

[a]For those with ≥ 1 corresponding healthcare encounter.

- On the WPAI, mean activity impairment was 40.7% (SD, 28.9%)
 - For employed patients, mean work productivity loss was 31.8%, presenteeism 27.3%, and absenteeism 7.8%
- Of the 13 patients who reported having a caregiver, the most common relationship was parent (46.2%), spouse, or significant other (23.1%)
 - 46.2% reported caregivers taking time off from work to help them
 - Most common caregiver activities were transportation for care and help with household duties (76.9% each), and assistance in treatment decisions or picking up medications (61.5% each)

Figure 2. Patient-reported impact of TDT on employment decisions

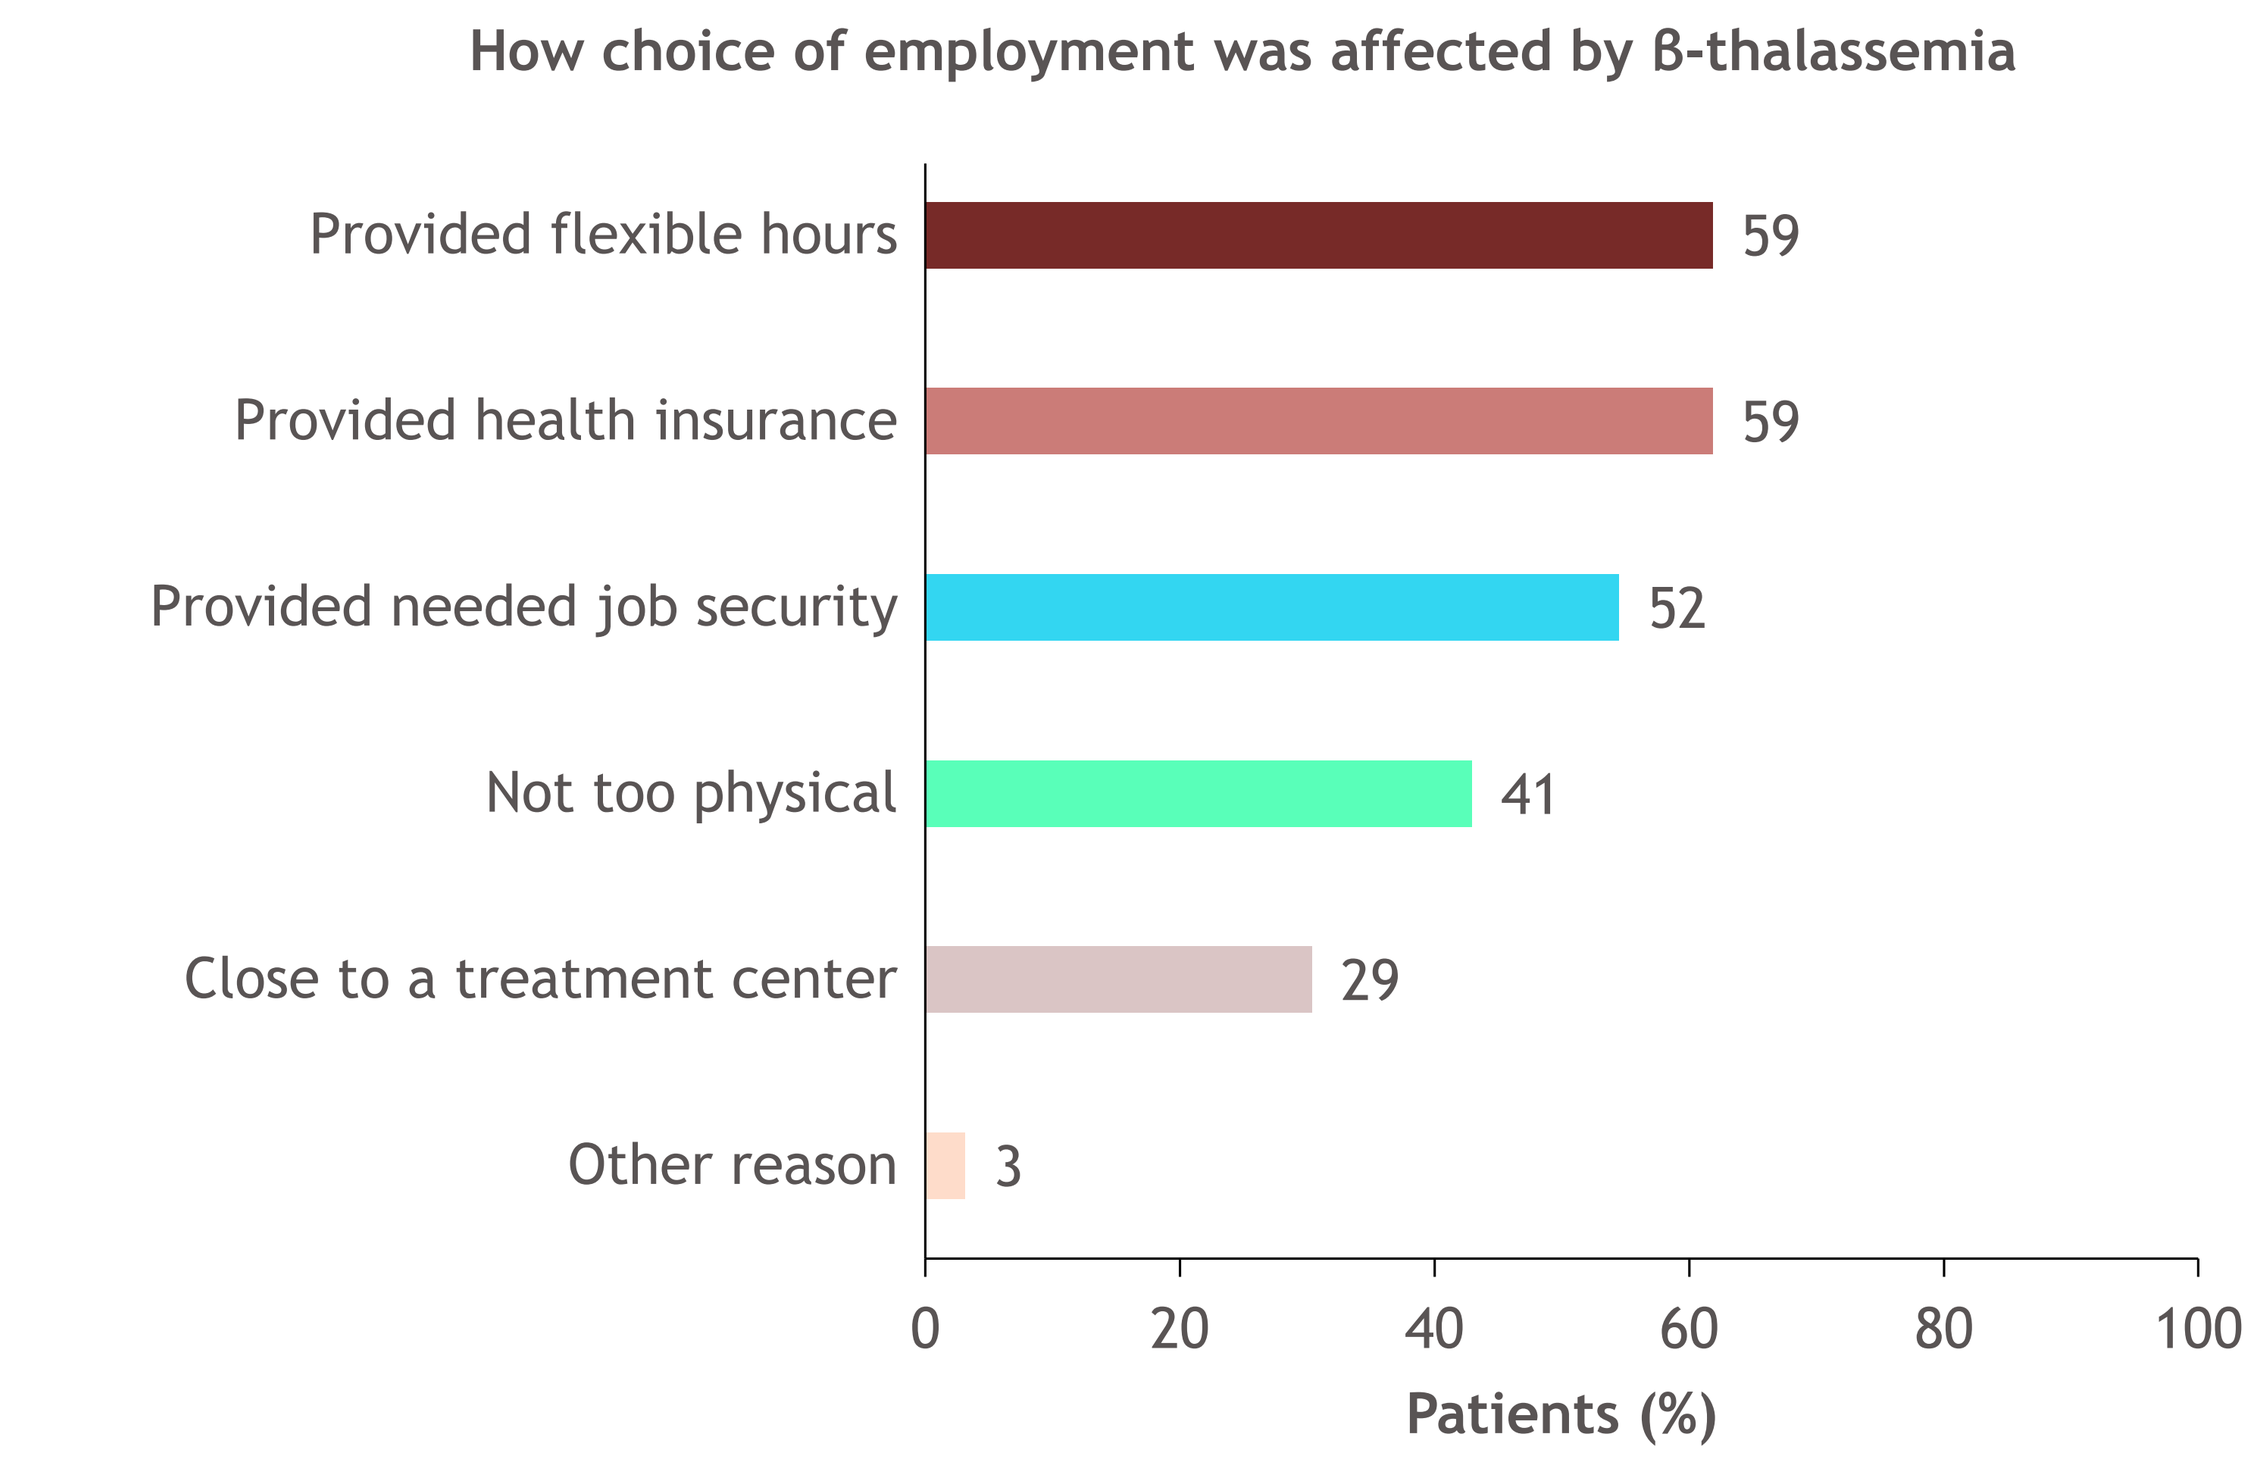
How choice of employment was affected by ß-thalassemia
Provided flexible hours
59
Provided health insurance
59
Provided needed job security
52
Not too physical
41
Close to a treatment center
29
Other reason
3
0
20
40
60
80
100
Patients (%)

Conclusions

- Patients undergoing chronic RBCT for TDT bear a substantial treatment-related HCRU burden, and incur considerable direct and indirect costs
- TDT impacts work productivity for patients and their caregivers
- Mean 6-month out-of-pocket costs related to care were USD 896.15
- Mean outside expenses for childcare, transportation, parking/tolls, medications, and other were USD 962.34 over 6 months
- RBCTs impose a meaningful economic burden to this predominantly working age, commercially insured population with TDT
- More effective treatment options that can reduce the frequency of RBCTs may help to reduce related HCRU and costs to patients and payers in the United States

References

1. Cappellini MD, et al. *Guidelines for the management of transfusion dependent thalassaemia*. 3rd ed. Nicosia, Cyprus: Thalassaemia International Federation; 2014.
2. Gan GG, et al. *Ann Acad Med Singap* 2016;45:520-523.
3. Scalone L, et al. *Curr Med Res Opin* 2008;24:1905-1917.
4. Pakbaz Z, et al. *Ann N Y Acad Sci* 2005;1054:457-461.
5. Price K, et al. *Blood* 2021;138(suppl 1). Abstract 4062.
6. Reilly Associates. WPAI General Information. http://www.reillyassociates.net/wpai_general.html. Published April 2019. Accessed March 14, 2022.

Acknowledgments

- The authors would like to thank Ali McBride, PharmD, of Bristol Myers Squibb for his valuable input on the presentation
- The study was supported by Bristol Myers Squibb
- All authors contributed to and approved the presentation; writing and editorial assistance were provided by Niamh Burke, PhD, of Excerpta Medica, funded by Bristol Myers Squibb

Disclosures

R.L.K.: Bristol Myers Squibb – employment, stock ownership. K.P., H.C., B.I., L.A., K.C., S.G.: Bristol Myers Squibb – consultancy, research funding; Cerner Enviza – employment.

Presented at the International Society for Pharmacoeconomics and Outcomes Research Annual Meeting; May 15-18, 2022; National Habor, MD, USA.

Email: Russell.Knoth@bms.com