Timely Albumin Infusion May Reduce Intensive Care Unit (ICU) Cost in Cirrhotic Patients with Spontaneous Bacterial Peritonitis (SBP): A Cross Sectional Study

M.C. Runken[1]; W.R. Kim[2]; K. Raghunathan[3]; G.S. Martin[4]; E. Viayna[5]; K. Lodaya[6]

[1]Grifols SSNA, Research Triangle Park, Durham, NC; [2]Division of Gastroenterology and Hepatology, Department of Medicine, Stanford University, Stanford, CA; [3]Department of Anesthesiology, Duke University Medical Center, Durham, NC; [4]Division of Pulmonary, Allergy, Critical Care and Sleep Medicine, Department of Medicine, Emory University School of Medicine and Grady Memorial Hospital, Atlanta, GA; [5]Grifols, S.A., Sant Cugat del Vallès, Spain; [6]Boston Strategic Partners, Inc., Boston, MA

BACKGROUND

- Spontaneous bacterial peritonitis is an infection of the ascites without an obvious intra-abdominal source, and is one of the most common and life-threatening complications of cirrhosis[1,2]
- In the US, complications related to SBP are associated with an 18% increase in the cost of treating decompensated cirrhosis[1]
- Albumin is the most abundant protein in plasma and plays a crucial role in oncotic pressure, substance transport and disposal, inflammation, oxidative stress, and endothelial stabilization
- Albumin adjunctive to antibiotics is more clinically effective in patients with SBP than antibiotics alone[3]
- Use of adjunctive albumin for SBP secondary to decompensated cirrhosis is more cost effective than antibiotics alone[3]
- American Association for the Study of Liver Diseases (AASLD) guidelines recommend timely administration of albumin in addition to antibiotics for SBP[4]

OBJECTIVE

- Using a real-world US claims database we sought to examine the relationship between albumin infusion timing and total ICU cost among cirrhotic inpatients who developed SBP during hospitalization

METHODS

- Premier Healthcare Database was queried for inpatient data corresponding to adult cirrhotic patients who developed SBP, received antibiotics, and experienced an ICU admission/transfer, between Jan-2016 and Jun-2019
- Patients were selected based on ICD-10 and billing codes
- The 'timely albumin' group included patients who received albumin within 1 day of hospitalization, while the 'non-timely' group received albumin after day 1 of hospitalization or not at all. Propensity score matching (PSM) (1:1) was used to balance both cohorts by sociodemographic characteristics, baseline Elixhauser Comorbidity Index, and presence of hepatorenal syndrome, end-stage renal disease, and ascites
- Generalized linear models were employed to determine the association between timely albumin and ICU costs, controlling for common comorbidities (i.e., hepatic encephalopathy)

Figure 1. Consort Diagram

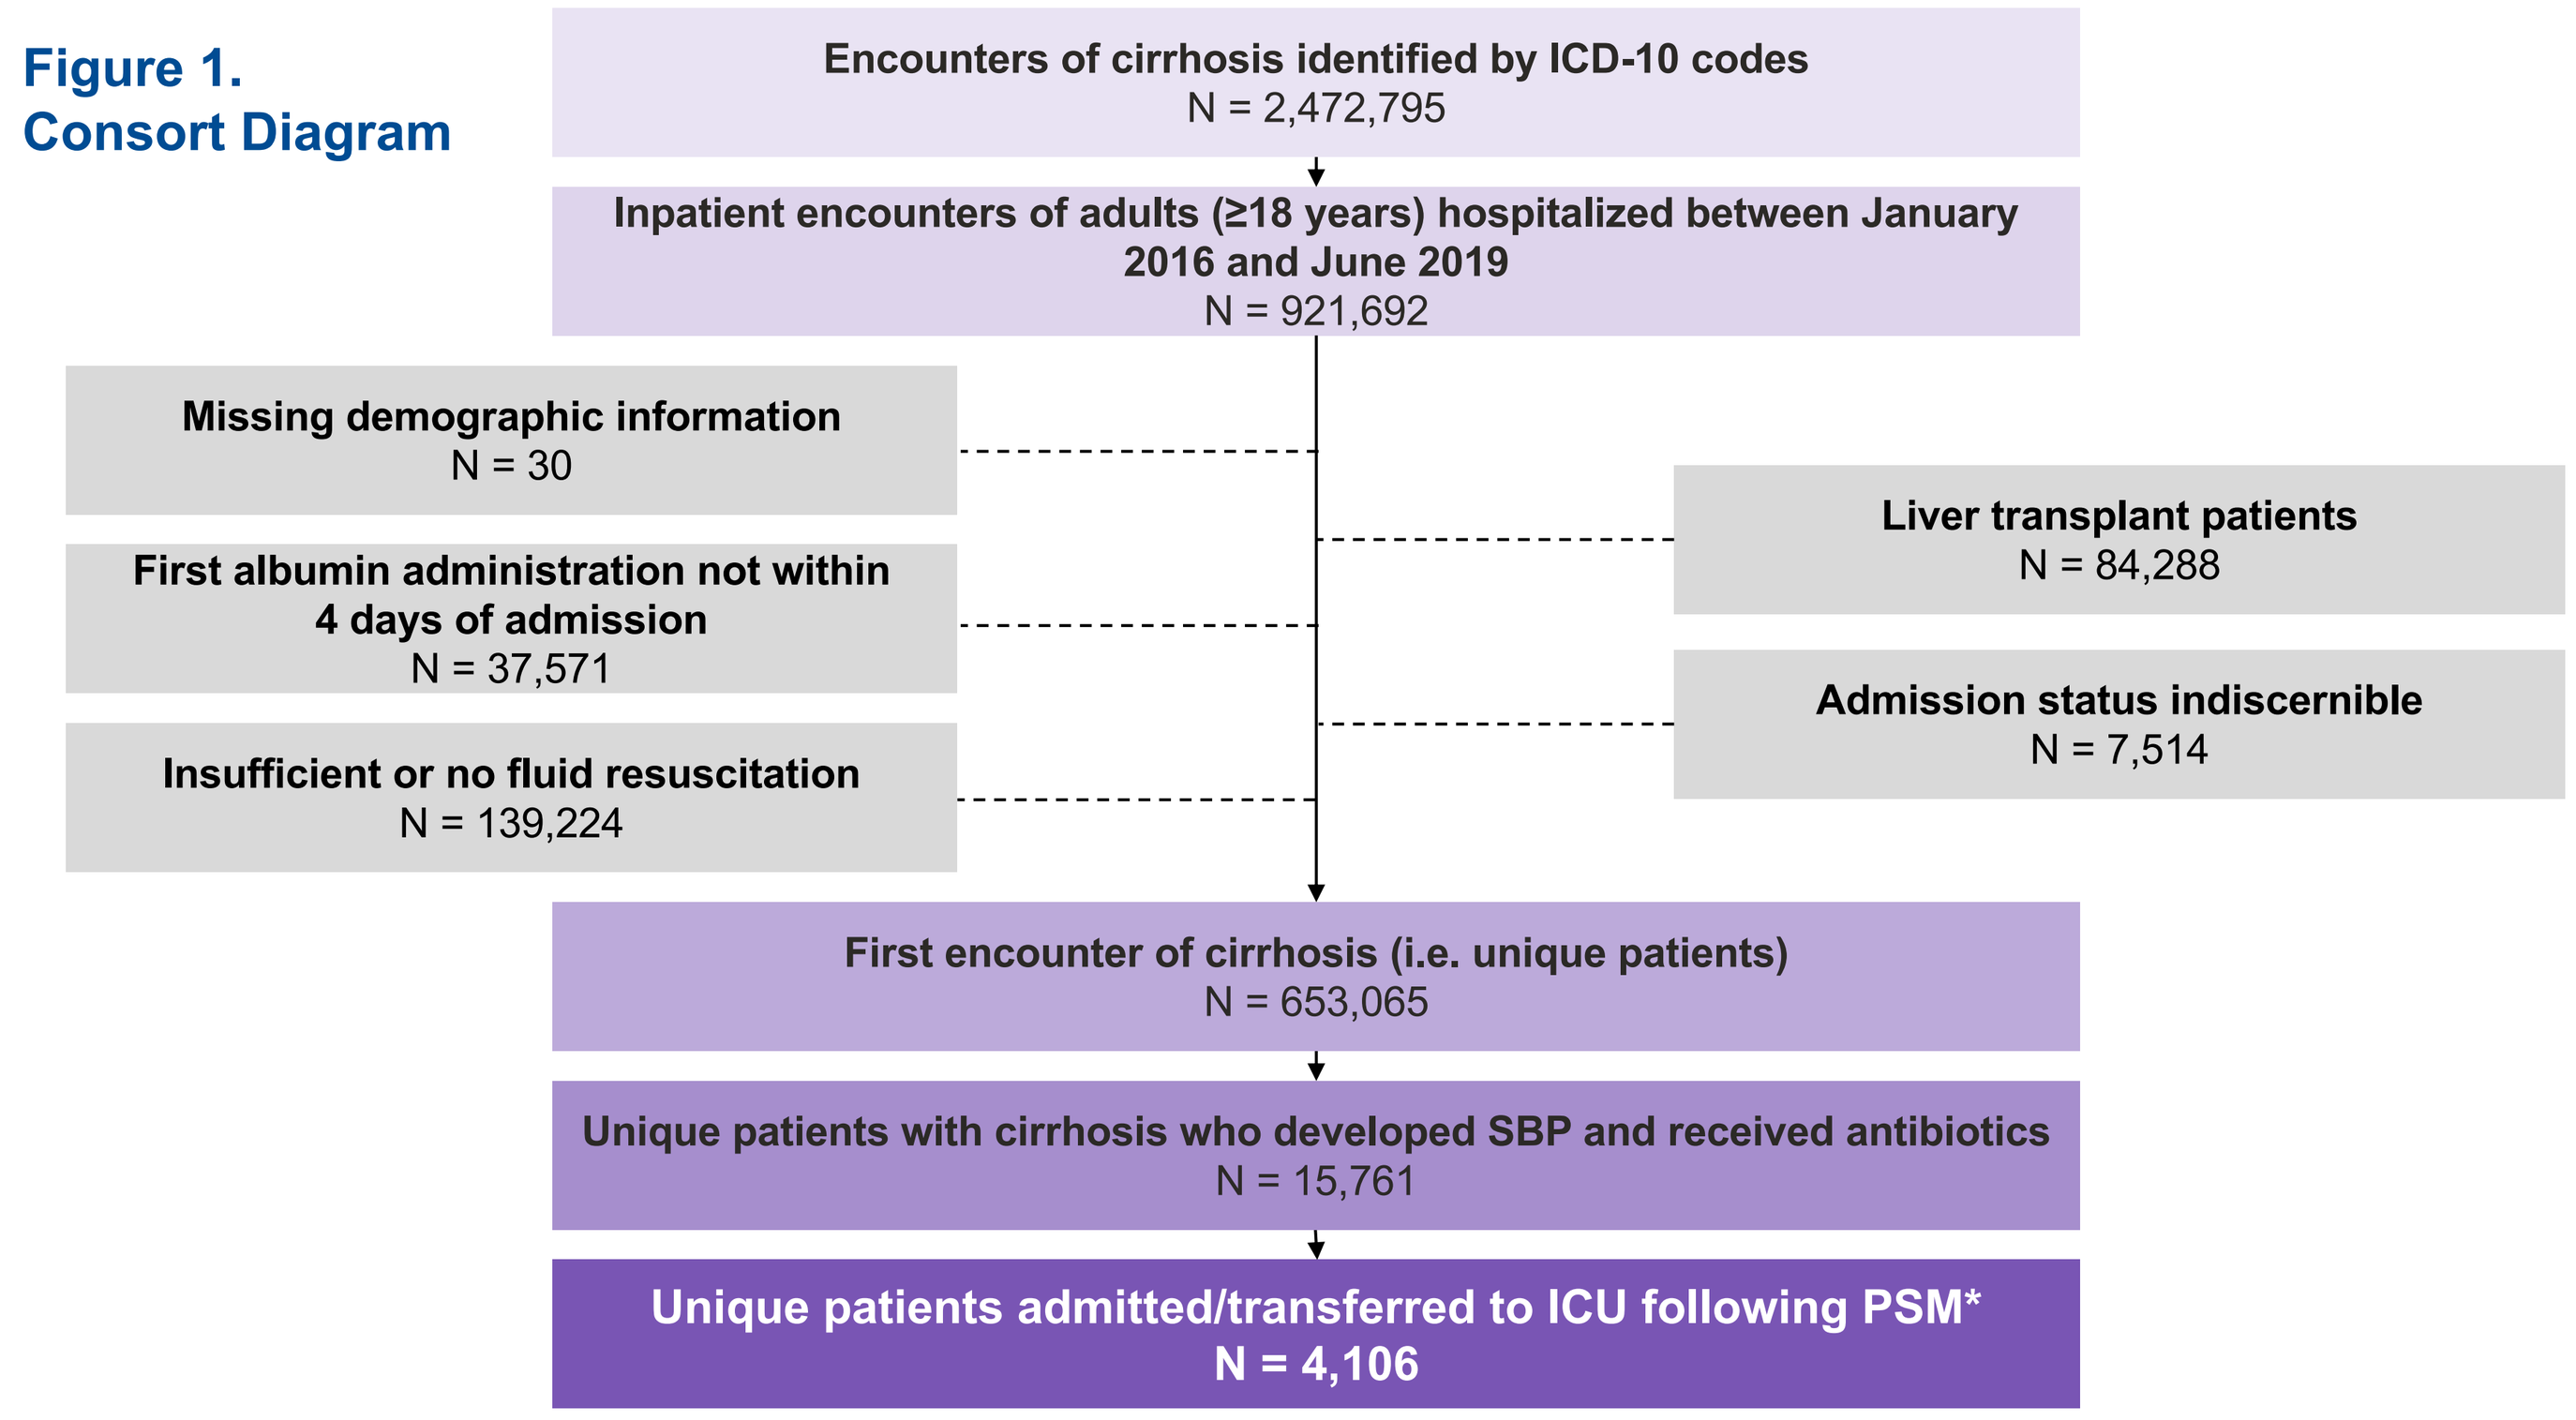
Encounters of cirrhosis identified by ICD-10 codes
N = 2,472,795
Inpatient encounters of adults (≥18 years) hospitalized between January 2016 and June 2019
N = 921,692
Missing demographic information
N = 30
First albumin administration not within 4 days of admission
N = 37,571
Insufficient or no fluid resuscitation
N = 139,224
Liver transplant patients
N = 84,288
Admission status indiscernible
N = 7,514
First encounter of cirrhosis (i.e. unique patients)
N = 653,065
Unique patients with cirrhosis who developed SBP and received antibiotics
N = 15,761
Unique patients admitted/transferred to ICU following PSM*
N = 4,106

RESULTS

- PSM yielded 4,106 total patients of whom 2,632 survived to discharge
- The risk-adjusted ICU cost ratio (CR) was 11% lower among all patients who received timely albumin compared to the non-timely group (CR: 0.89, 95% CI: 0.84-0.93, p<0.001)
- In the survivor cohort, the risk-adjusted ICU CR was 18% lower in the timely group relative to the non-timely group (CR: 0.82, 95% CI: 0.77-0.88, p<0.001)

Table 1. Patient Demographics and Hospital Characteristics

	ALL PATIENTS N = 4,106 visits		SURVIVORS ONLY N = 2,632 visits	
	Timely Albumin	Non-Timely Albumin	Timely Albumin	Non-Timely Albumin
	N = 2,053 (50%)	N = 2,053 (50%)	N = 1,316 visits (50%)	N = 1,316 visits (50%)
Characteristic				
Sex				
Male	1276 (62.2%)	1268 (61.8%)	820 (62.3%)	832 (63.2%)
Female	777 (37.8%)	785 (38.2%)	496 (37.7%)	484 (36.8%)
Age (years)				
18-29	26 (1.3%)	28 (1.4%)	21 (1.6%)	25 (1.9%)
30-49	427 (20.8%)	421 (20.5%)	274 (20.8%)	286 (21.7%)
50-64	988 (48.1%)	1005 (49.0%)	612 (46.5%)	584 (44.4%)
65+	612 (29.8%)	599 (29.2%)	409 (31.1%)	421 (32.0%)
Ethnicity				
White	1552 (75.6%)	1559 (75.9%)	1008 (76.6%)	1001 (76.1%)
Black	156 (7.6%)	163 (7.9%)	88 (6.7%)	99 (7.5%)
Other	309 (15.1%)	297 (14.5%)	196 (14.9%)	190 (14.4%)
Not Specified	36 (1.8%)	34 (1.7%)	24 (1.8%)	26 (2.0%)
Admission Type				
Elective	62 (3.0%)	57 (2.8%)	39 (3.0%)	41 (3.1%)
Emergency	1799 (87.6%)	1797 (87.5%)	1154 (87.7%)	1152 (87.5%)
Trauma	12 (0.6%)	12 (0.6%)	8 (0.6%)	6 (0.5%)
Urgent	180 (8.8%)	187 (9.1%)	115 (8.7%)	117 (8.9%)
Payor Group				
Medicare	840 (40.9%)	831 (40.5%)	567 (43.1%)	559 (42.5%)
Managed Care, Commercial	466 (22.7%)	467 (22.7%)	296 (22.5%)	292 (22.2%)
Medicaid	564 (27.5%)	563 (27.4%)	351 (26.7%)	355 (27.0%)
Self Pay	97 (4.7%)	103 (5.0%)	53 (4.0%)	59 (4.5%)
Other	86 (4.2%)	89 (4.3%)	49 (3.7%)	51 (3.9%)
Hospital Characteristics at Index Visit				
Hospital Type				
Urban	1835 (89.4%)	1812 (88.3%)	1166 (88.6%)	1169 (88.8%)
Rural	218 (10.6%)	241 (11.7%)	150 (11.4%)	147 (11.2%)
Teaching Facility				
Yes	1141 (55.6%)	1147 (55.9%)	694 (52.7%)	687 (52.2%)
No	912 (44.4%)	906 (44.1%)	622 (47.3%)	629 (47.8%)
Clinical Characteristics and Conditions At/Before Index Visit				
Baseline Elixhauser Comorbidity Index (ECI), Mean ± SD	21.0 ± 8.9	21.1 ± 9.5	20.4 ± 8.9	20.6 ± 9.4
ECI, Median [25th, 75th]	21.0 [16.0, 27.0]	21.0 [15.0, 27.0]	20.0 [15.0, 26.0]	20.0 [14.0, 27.0]
Baseline Hepatorenal Syndrome	715 (34.8%)	648 (31.6%)	400 (30.4%)	360 (27.4%)
Baseline ESRD	285 (13.9%)	298 (14.5%)	181 (13.8%)	173 (13.1%)
Baseline Ascites	1238 (60.3%)	1221 (59.5%)	817 (62.1%)	803 (61.0%)
Acute Kidney Injury (AKI)	1727 (84.1%)	1570 (76.5%)	1047 (79.6%)	927 (70.4%)
Hepatic Encephalopathy (HE)	799 (38.9%)	666 (32.4%)	501 (38.1%)	411 (31.2%)
Glucose Level				
Both Hyper- and Hypoglycemia	433 (21.1%)	432 (21.0%)	231 (17.6%)	242 (18.4%)
Hyperglycemia only	500 (24.4%)	510 (24.8%)	356 (27.1%)	345 (26.2%)
Hypoglycemia only	461 (22.5%)	456 (22.2%)	256 (19.5%)	246 (18.7%)
Neither Hyper- nor hypoglycemia	659 (32.1%)	655 (31.9%)	473 (35.9%)	483 (36.7%)
Exposures/Clinical Interventions During Index Visit				
Large Volume Paracentesis (LVP)	1323 (64.4%)	1183 (57.6%)	873 (66.3%)	755 (57.4%)
GI Bleeding (GIB) and Transfusions				
Both	243 (11.8%)	305 (14.9%)	134 (10.2%)	189 (14.4%)
GIB only	123 (6.0%)	127 (6.2%)	78 (5.9%)	74 (5.6%)
Transfusions only	771 (37.6%)	718 (35.0%)	484 (36.8%)	440 (33.4%)
Neither	916 (44.6%)	903 (44.0%)	620 (47.1%)	613 (46.6%)
Use of Mechanical Ventilation	707 (34.4%)	747 (36.4%)	259 (19.7%)	316 (24.0%)
Vasopressor Use	1381 (67.3%)	1151 (56.1%)	732 (55.6%)	571 (43.4%)
Use of Diuretics	1316 (64.1%)	1419 (69.1%)	899 (68.3%)	955 (72.6%)
Use of Steroids	367 (17.9%)	466 (22.7%)	227 (17.2%)	300 (22.8%)
Use of Beta Blockers	289 (14.1%)	407 (19.8%)	229 (17.4%)	322 (24.5%)

SUMMARY/CONCLUSION

Timely albumin infusion was associated with a significant ICU cost reduction –

suggesting that the co-administration of albumin with antibiotics may alleviate the growing ICU cost burden of SBP as a complication of cirrhosis.

Figure 3: Significant ICU Cost Saving Among Patients Treated with Albumin in a Timely Manner

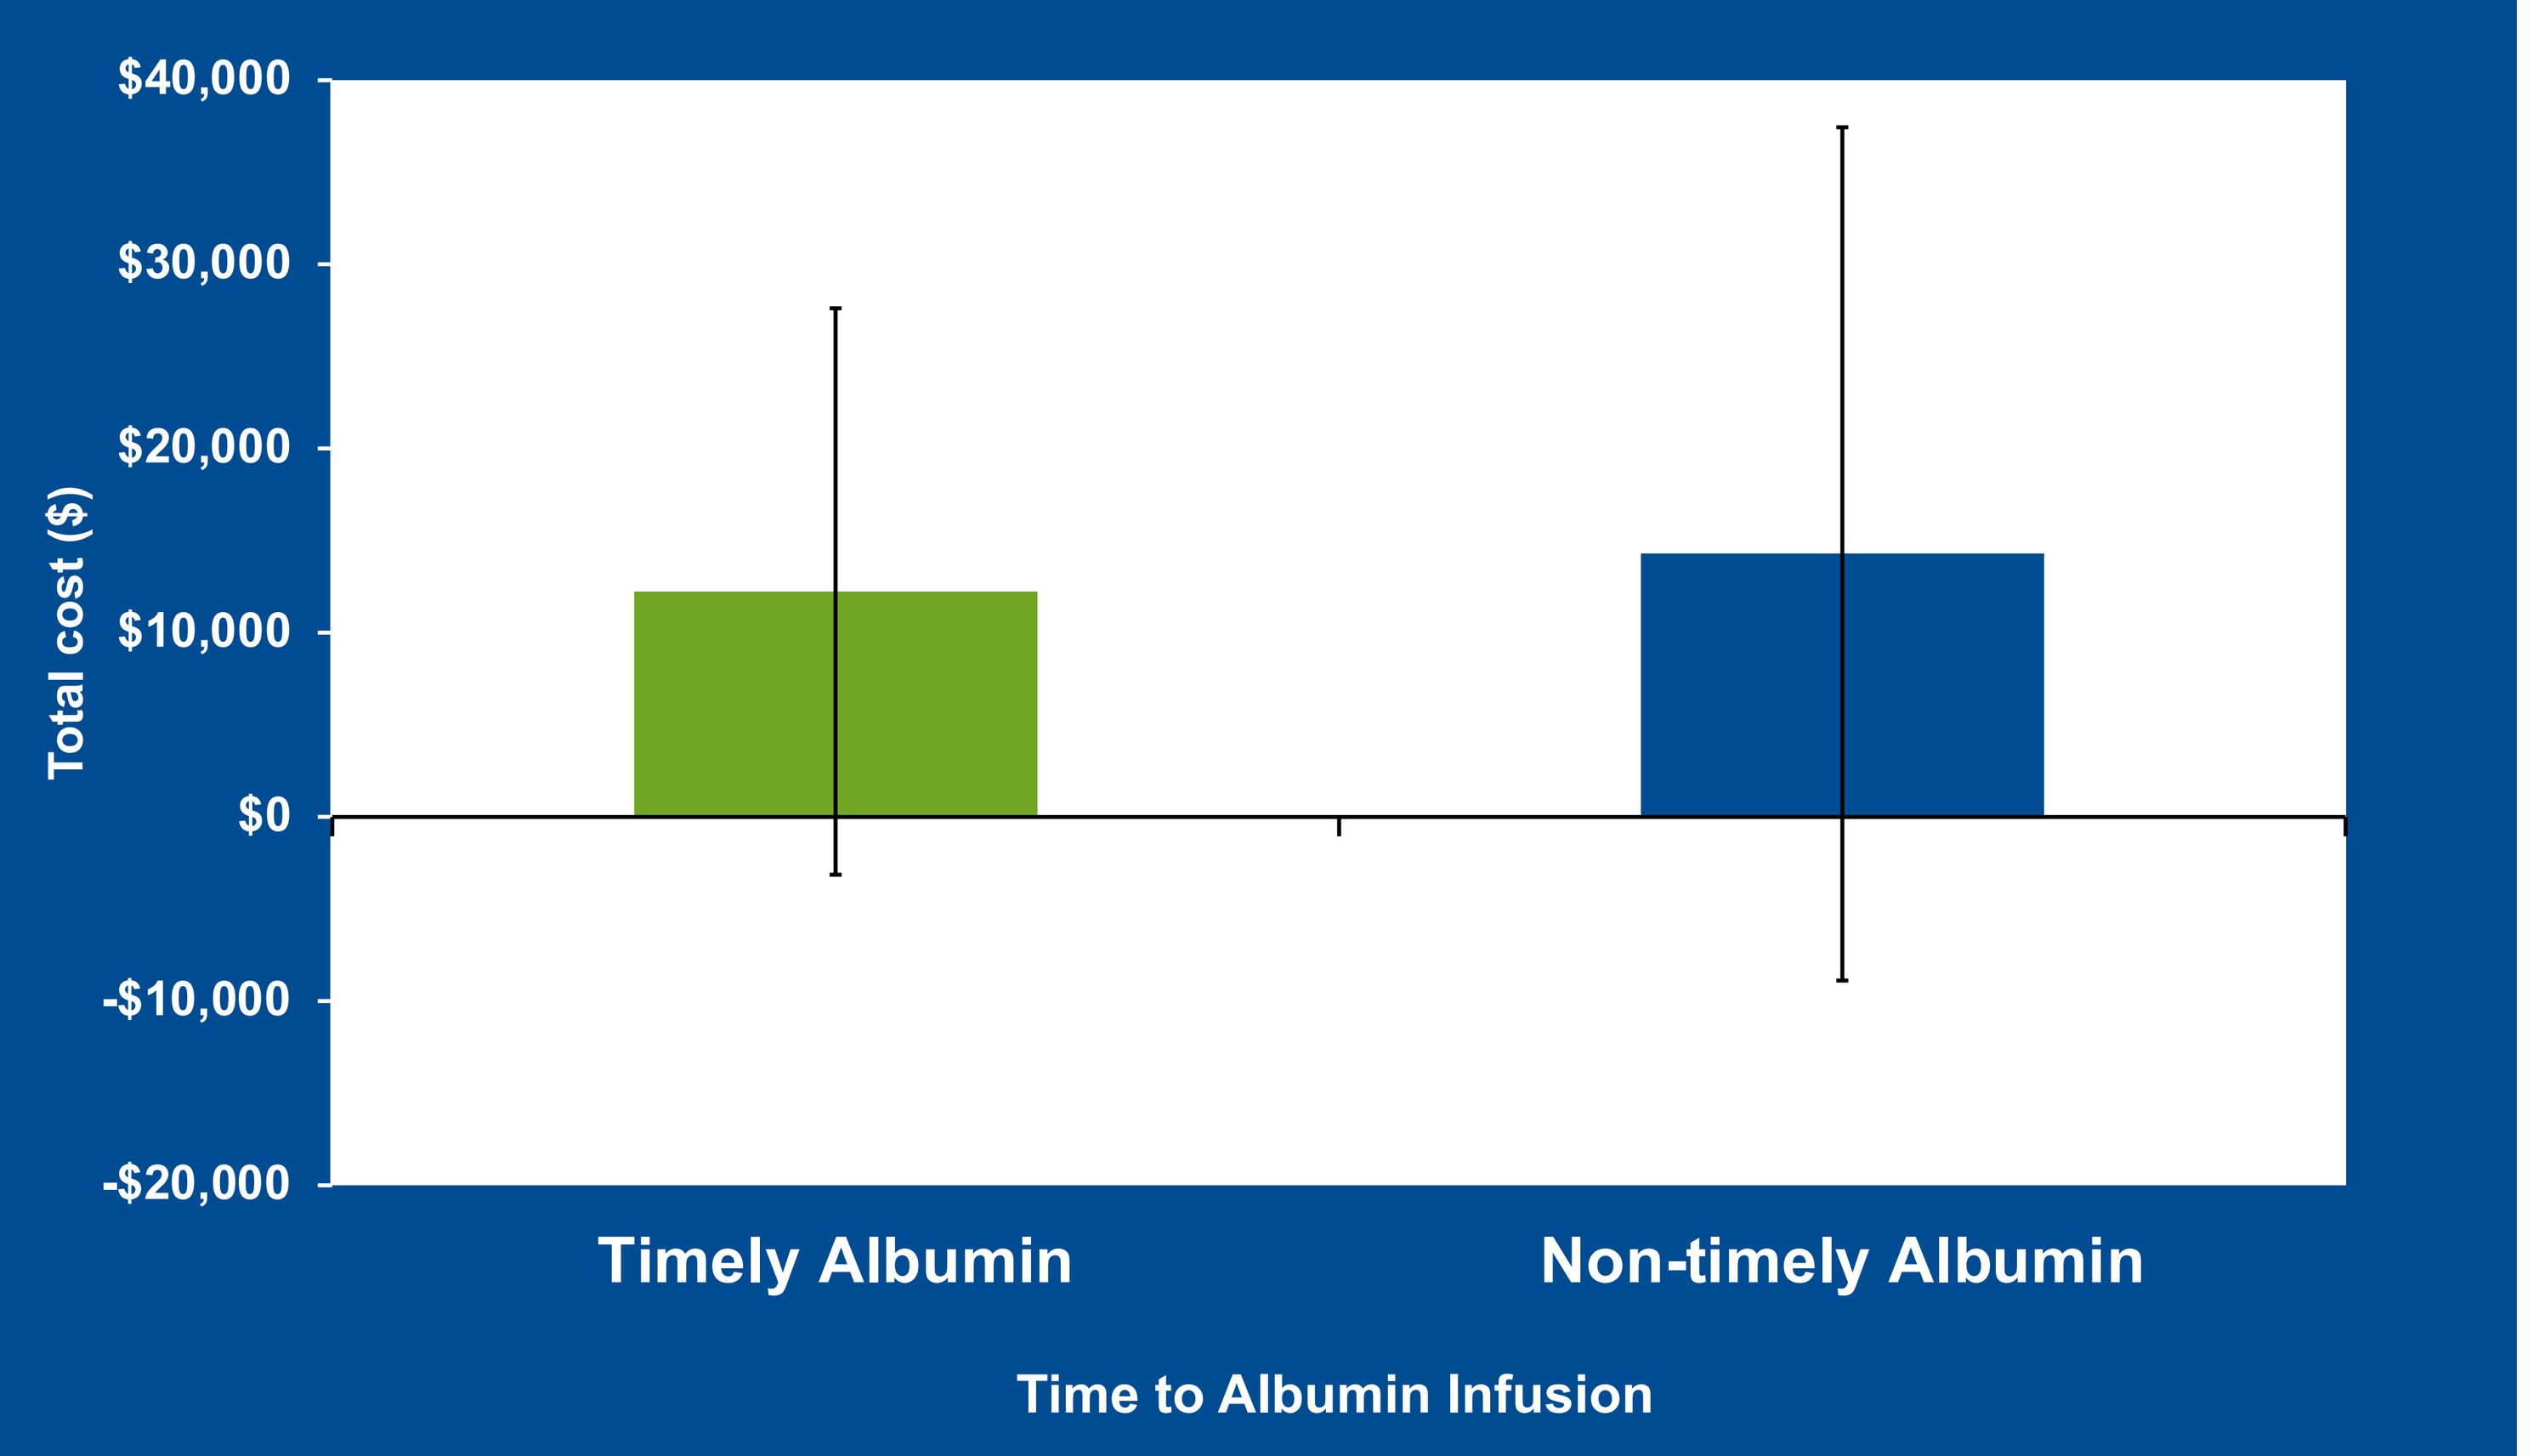
$40,000
$30,000
$20,000
$10,000
$0
-$10,000
-$20,000
Total cost ($)
Timely Albumin
Non-timely Albumin
Time to Albumin Infusion

Table 2: Association Between Timely Albumin and Total Care Costs after 1:1 Propensity Score Matching (PSM)

All Patients (N = 4,106) Surviving Patients (N = 2,632)	Odds Ratio (OR)		95% Confidence Interval (CI) Lower Limit		95% Confidence Interval (CI) Upper Limit	
Characteristic	All Patients	Surviving Patients	All Patients	Surviving Patients	All Patients	Surviving Patients
Timely (≤1 day of Hospital Admission) vs. Non-Timely (None or Late) Albumin Administration	0.89*	0.82*	0.84	0.77	0.93	0.88
Acute Kidney Injury (AKI)	1.05	1.04	0.98	0.97	1.12	1.12
Large Volume Paracentesis (LVP)	1.13*	1.14*	1.08	1.07	1.19	1.21
Hepatic Encephalopathy (HE)	1.02	1.07*	0.97	1.00	1.08	1.13
Gastrointestinal Bleeding (GIB) and Transfusions						
Both	1.30*	1.06	1.20	0.96	1.40	1.17
GIB only	1.23*	1.11	1.11	0.97	1.37	1.29
Transfusions only	1.28*	1.21*	1.21	1.13	1.36	1.47
Mechanical Ventilation	1.86*	2.12*	1.75	1.96	1.97	2.29
Vasopressors**	1.52*	1.56*	1.44	1.46	1.61	1.66
Glucose Level						
Both	1.63*	1.51*	1.51	1.38	1.75	1.65
Hyperglycemia	1.26*	1.14*	1.18	1.06	1.35	1.23
Hypoglycemia	1.09*	1.12*	1.01	1.03	1.16	1.22
Diuretics	1.19*	1.16*	1.12	1.09	1.25	1.24
Steroids***	1.24*	1.06	1.16	0.99	1.32	1.15
Beta Blockers****	0.91*	0.89*	0.85	0.83	0.97	0.96

*p < 0.05; **Vasopressors identified included: dobutamine, dopamine, epinephrine, norepinephrine, vasopressin; ***Steroids included budesonide, cortisone, dexamethasone, dexamethasone/lidocaine, fludrocortisone, methylprednisolone, prednisolone, prednisone; ****Beta blockers included carvedilol, nadolol, nadolol/Bendroflumethiazide, propranolol, propranolol/ hydrochlorothiazide

Figure 2: Cost Ratio Forest Plot

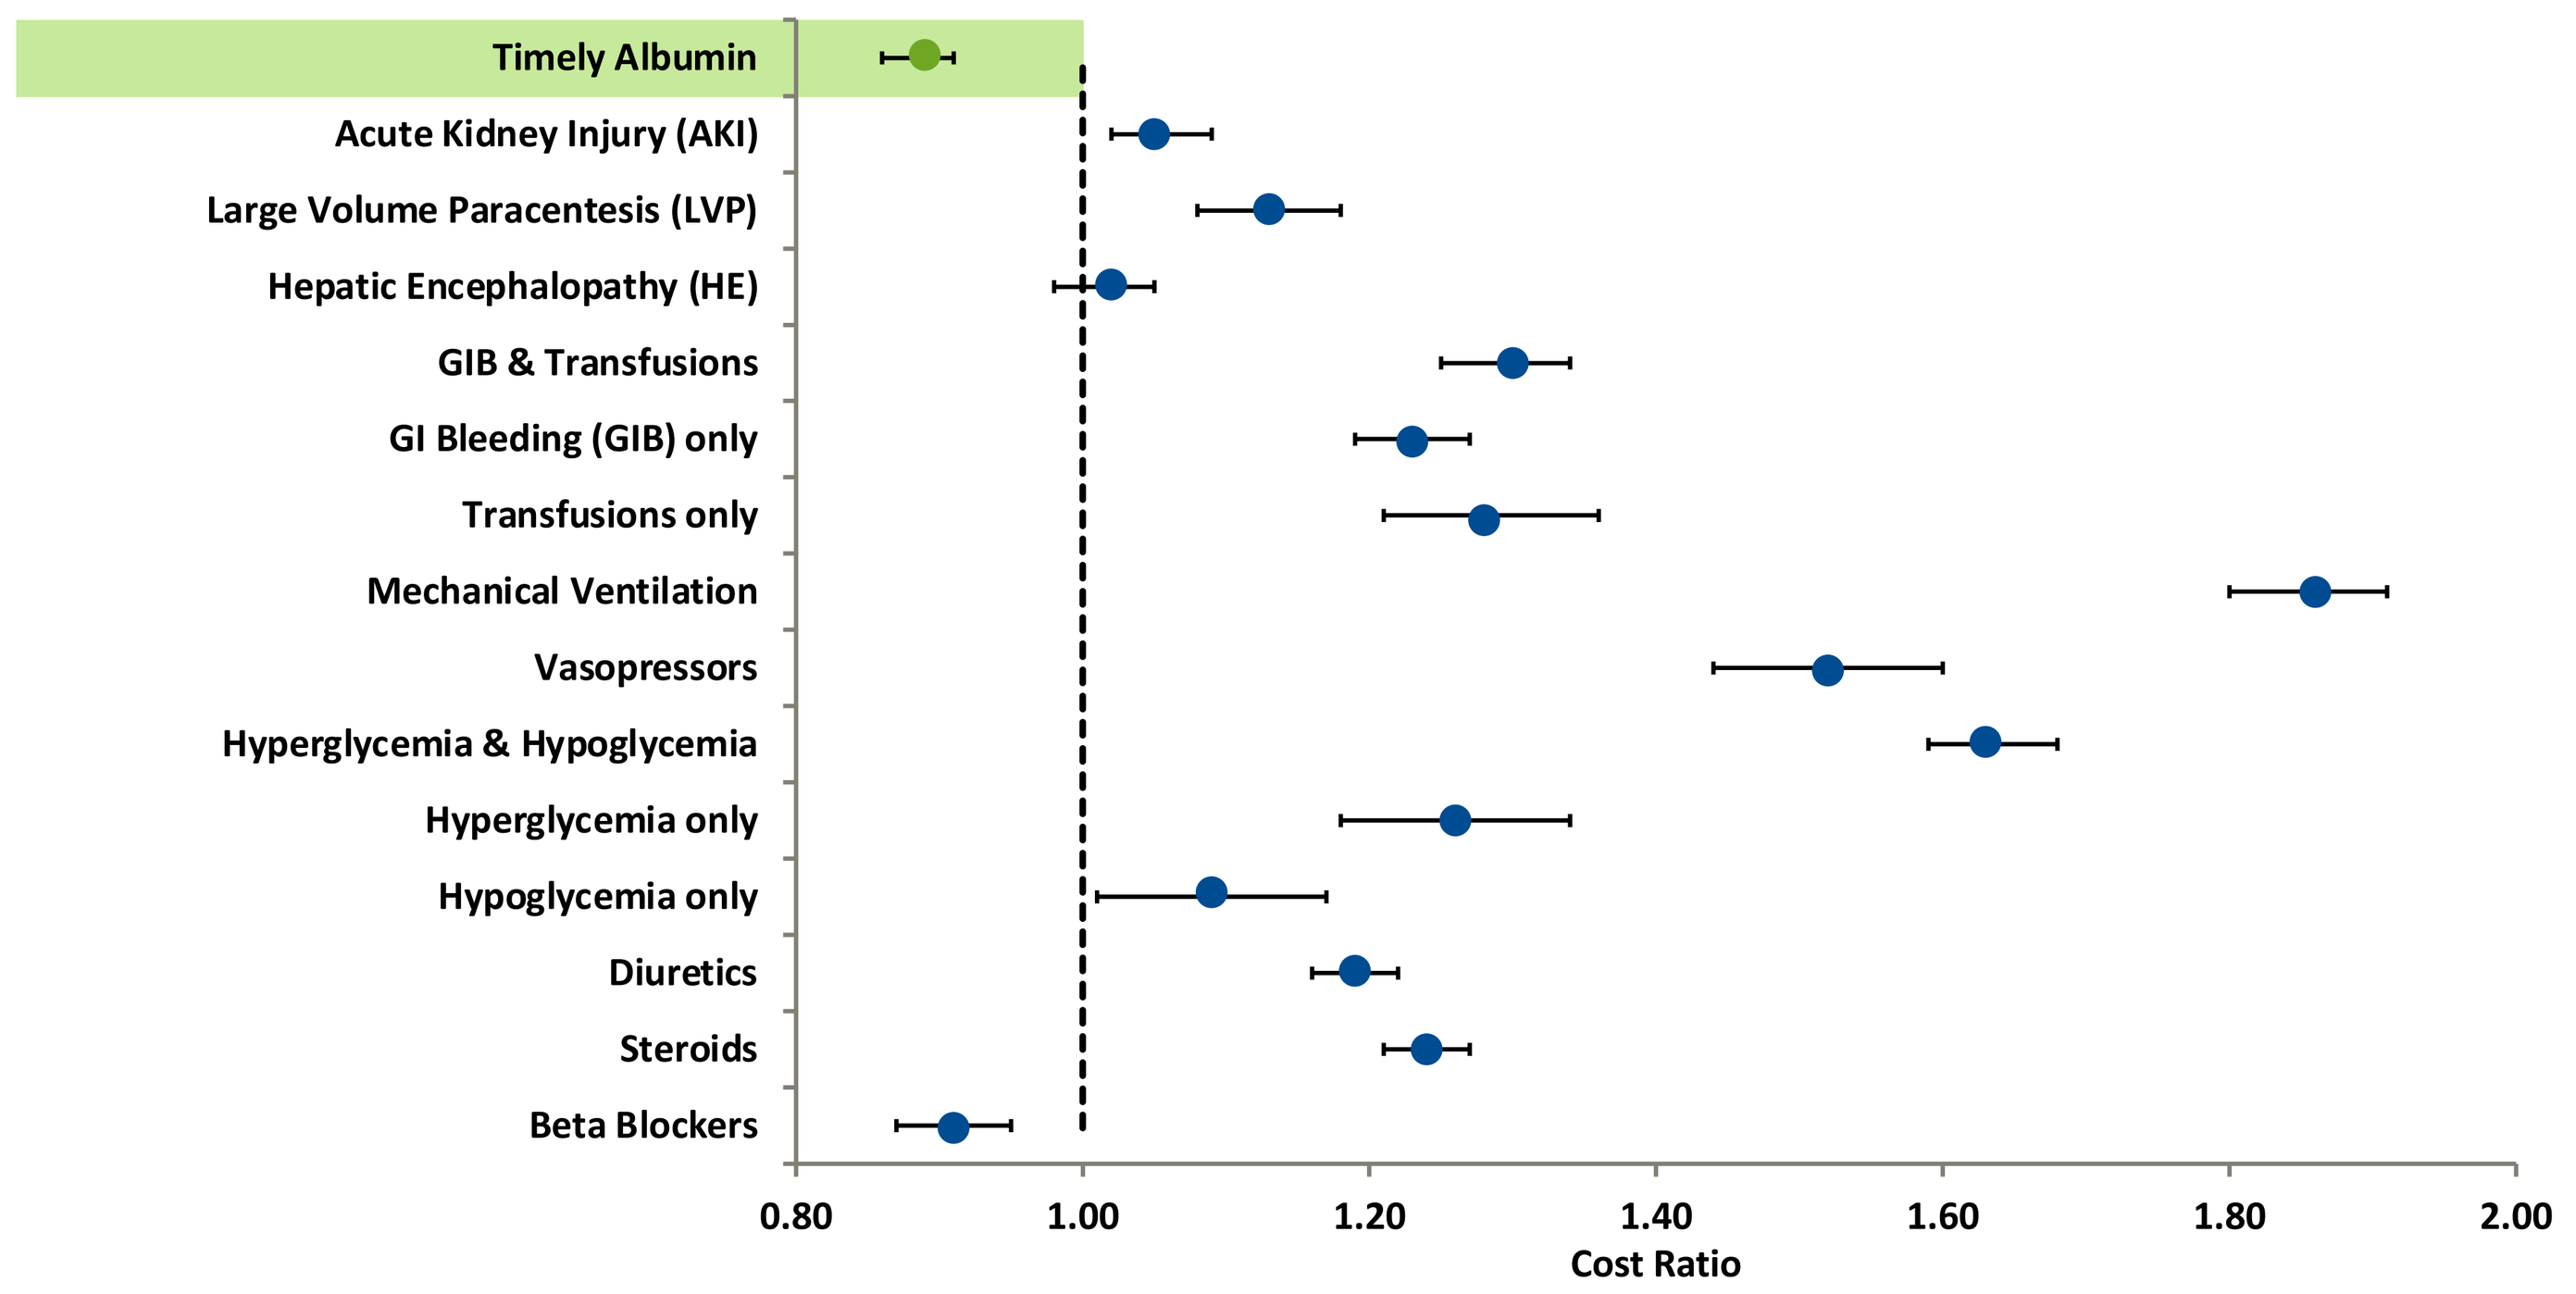
Timely Albumin
Acute Kidney Injury (AKI)
Large Volume Paracentesis (LVP)
Hepatic Encephalopathy (HE)
GIB & Transfusions
GI Bleeding (GIB) only
Transfusions only
Mechanical Ventilation
Vasopressors
Hyperglycemia & Hypoglycemia
Hyperglycemia only
Hypoglycemia only
Diuretics
Steroids
Beta Blockers
0.80
1.00
1.20
1.40
1.60
1.80
2.00
Cost Ratio

REFERENCES:

1. Pleguezuelo, M., Benitez, J. M., Jurado, J., Montero, J. L. & De la Mata, M. Diagnosis and management of bacterial infections in decompensated cirrhosis. *World J. Hepatol*. 5, 16–25 (2013).
2. Koulaouzidis, A., Bhat, S., Karagiannidis, A., Tan, W. C. & Linaker, B. D. Spontaneous bacterial peritonitis. Postgrad. *Med. J.* 83, 379–383 (2007)
3. Runken, M. C. et al. The cost-effectiveness of albumin in the treatment of decompensated cirrhosis in Germany, Italy, and Spain. Health Econ. Rev. 9, 22 (2019).
4. Biggins, S. W. et al. Diagnosis, Evaluation, and Management of Ascites, Spontaneous Bacterial Peritonitis and Hepatorenal Syndrome: 2021 Practice Guidance by the American Association for the Study of Liver Diseases. *Hepatology* 74, 1014–1048 (2021).

Presented at ISPOR 2022 | May 15-18, 2022 | Washington, DC and Virtual
Acknowledgement: This study was funded by Grifols SSNA (Research Triangle Park, NC, USA).

GRIFOLS