Treatment patterns in metastatic urothelial cancer (mUC): A real-world survey in France, Germany, Italy, Spain, and the UK (Eu5)

N. Milloy,[1] M. Kirker,[2] M. Berry,[1] R. Montgomery,[1] C. Bleasdale,[1] M. Kearney,[3] N. Costa,[4] J. Chang[2]

[1]Adelphi Real World, Bollington, UK; [2]Pfizer, New York, NY, USA; [3]the healthcare business of Merck KGaA, Darmstadt, Germany; [4]Pfizer, Porto Salvo, Portugal

SCOPE

- The aim of this study was to understand mUC treatment patterns in the Eu5
- Patient demographics, clinical characteristics, and treatment patterns among first-line (1L), second-line (2L) and third-line (3L) patients with mUC were assessed
- Responses to 1L platinum-based chemotherapy were evaluated

CONCLUSIONS

- Platinum-based chemotherapy is the 1L standard of care for patients with mUC, consistent with recommendations by European Association of Urology (EAU)[1] and European Society for Medical Oncology (ESMO) guidelines[2]
- Among all patients who completed 1L platinum-based chemotherapy and initiated 2L treatment, the most common type of response was a partial response
- ICI monotherapy was the most common 2L treatment received, and best supportive care was the most common at 3L
- Following the European Commission's approval in January 2021 of avelumab for 1L maintenance treatment in patients who did not progress following platinum-based chemotherapy,[3] future studies should continue to evaluate treatment patterns for patients with mUC and assess patient outcomes as the treatment landscape evolves

GET POSTER PDF

Copies of this poster obtained through this quick response (QR) code are for personal use only and may not be reproduced without permission from ISPOR and the author of this poster

Correspondence: **Melissa Kirker, Melissa.Kirker@pfizer.com**

BACKGROUND

- Urothelial cancers are the tenth most common cancer diagnosed worldwide[4]
 - The prognosis of metastatic disease is poor, with few approved therapeutic options
- ESMO and EAU guidelines recommend that 1L treatment selection for mUC should be based on platinum eligibility status[1,2]
- Platinum-based chemotherapy is the standard 1L treatment approach in eligible patients with mUC
 - In the 2L setting, ICIs are commonly used therapies

METHODS

Data source

- Real-world data were drawn from the Adelphi mUC Disease Specific Programme (DSP),[5] a real-world point-in-time study conducted between November 2020 and April 2021
- Oncologists and urologists completed patient record forms for their next 8 consecutive eligible patients with mUC who visited, which included collection of patients' demographics, clinical characteristics, and treatment patterns

Eligibility criteria

Physician inclusion criteria

- Specialty in medical oncology or urology
- Management of ≥8 patients with mUC per month
- Personal responsibility for prescribing decisions

Patient inclusion criteria

- Age ≥18 years
- Confirmed diagnosis of mUC
- No involvement in a clinical trial at time of data collection

Ethics

- This research obtained an ethics exemption from the Western Institutional Review Board

Statistical analysis

- Descriptive analyses were conducted, and any missing data were excluded; statistical comparisons were not conducted

Addition requirements

- Quotas of three 1L, two 2L, and three 3L+ patients were implemented

RESULTS

- A total of 232 oncologists and urologists provided data from 1,922 patients with mUC across the Eu5 (**Table 1**)

Patient characteristics

- Patients were 73% male and 27% female (**Table 1**)
- The mean (SD) age at diagnosis was 69 (7.9) years, and was similar across all countries
- The primary tumor was the located in the lower urinary tract in 83% of patients
- Patients started 1L treatment at a mean (SD) of 1.6 (3.5) months after mUC diagnosis
- At initial diagnosis of mUC, 81% of patients had ≥2 site of metastasis
- Metastases were most commonly located in the lymph nodes (58%)
- ECOG PS was 0-1 in 84% of patients at diagnosis of mUC

Treatment patterns

- Treatments patterns described per line of therapy were either ongoing or completed (**Figure 1**)
- Among patients who received 1L treatment in the Eu5 (n=1,922):
 - Platinum-based chemotherapy was used in 82% of patients, ranging from 77% to 85% by country
 - ICI monotherapy was administered to 10% of patients
- Among patients who received 2L in the Eu5 (n=1,177):
 - ICI was the most common treatment received (63%), with use varying from 59% in Italy to 66% in France
 - 2L chemotherapy use was highest in Italy (37%) and lowest in France (30%)
- Among patients who received 3L in the Eu5 (n=627):
 - Best supportive care was received in 41% of patients, ranging from 23% in Spain, 35% in France, 40% in Italy, 48% in Germany, and 57% in the UK
 - Non–platinum-based chemotherapy was received by 36% of patients, and 17% received an ICI
- At the time of data collection, 980 patients had completed 1L platinum-based chemotherapy and had initiated 2L treatment; 82% of those patients achieved disease control
 - Partial response to platinum chemotherapy was achieved in 64% of patients
 - Complete response was achieved in 14% of patients
 - Patients who completed 1L platinum-based chemotherapy and initiated 2L treatment received a median of 6.0 cycles of 1L platinum-based chemotherapy, whether cisplatin or carboplatin-based regimens

Limitations

- These data were collected via a point-in-time chart review conducted by physicians, and access to the source records was not provided. Although efforts were made to reduce inconsistences, missing data or discrepancies could not be verified with source chart information
- Although minimal inclusion criteria governed the selection of the participating physicians in order to provide a geographically diverse sample, participation was influenced by physician willingness to complete the survey
- Physicians were asked to provide data for a consecutive series of patients to avoid selection bias, but no formal patient selection verification procedures were in place
- The point-in-time design of the DSP survey prevents any conclusions about causal relationships in the analysis
- Recall bias, a common limitation of surveys, might have also affected physician responses to the DSP questionnaires

Table 1. Patient demographics

	Eu5 N=1,922	France n=465	Germany n=400	Italy n=360	Spain n=361	UK n=336
Sex, n (%)						
Male	1,405 (73)	356 (77)	308 (77)	244 (68)	272 (75)	225 (67)
Female	517 (27)	109 (23)	92 (23)	116 (32)	89 (25)	111 (33)
Age at time of data collection, years						
Mean (SD)	69.1 (7.9)	70.0 (8.1)	69.7 (7.3)	68.9 (8.2)	68.2 (7.8)	68.3 (7.9)
Treatment received to date, n (%)						
1L	745 (39)	197 (42)	148 (37)	134 (37)	138 (38)	128 (38)
2L	550 (29)	128 (28)	121 (30)	98 (27)	107 (30)	96 (29)
≥3L	627 (33)	140 (30)	131 (33)	128 (36)	116 (32)	112 (33)
Sites of metastases at diagnosis of mUC (top 5), n (%)						
Lymph nodes	1,110 (58)	283 (61)	184 (46)	240 (67)	215 (60)	188 (56)
Lungs	792 (41)	216 (46)	107 (27)	163 (45)	151 (42)	155 (46)
Bone	562 (29)	128 (28)	126 (32)	121 (34)	109 (30)	78 (23)
Liver	406 (21)	138 (30)	65 (16)	64 (18)	70 (19)	69 (21)
Adrenal glands	125 (7)	37 (8)	17 (4)	13 (4)	34 (9)	24 (7)
Time to 1L treatment initiation after diagnosis of mUC, months						
Mean (SD)	1.6 (3.5)	1.5 (3.2)	2.1 (4.3)	1.7 (3.7)	1.1 (2.2)	1.6 (3.5)
ECOG PS at diagnosis of mUC, n (%)						
0-1	1,618 (84)	353 (76)	329 (82)	329 (91)	298 (83)	309 (92)
2+	290 (15)	111 (24)	70 (18)	23 (6)	62 (17)	24 (7)
Unknown/not assessed	14 (1)	1 (<1)	1 (<1)	8 (2)	1 (<1)	3 (1)
Primary tumor location at initial diagnosis of UC, n (%)						
Upper urinary tract	321 (17)	55 (12)	69 (17)	81 (22)	65 (18)	51 (15)
Lower urinary tract	1,601 (83)	410 (88)	331 (83)	279 (78)	296 (82)	285 (85)

1L, first line; **2L**, second line; **3L**, third line; **ECOG PS**, Eastern Cooperative Oncology Group performance status; **Eu5**, France, Germany, Italy, Spain, and UK; **mUC**, metastatic urothelial cancer; **UC**, urothelial cancer.

Figure 1. mUC treatments received at 1L, 2L, and 3L in the Eu5

1L mUC treatment (n=1,922)

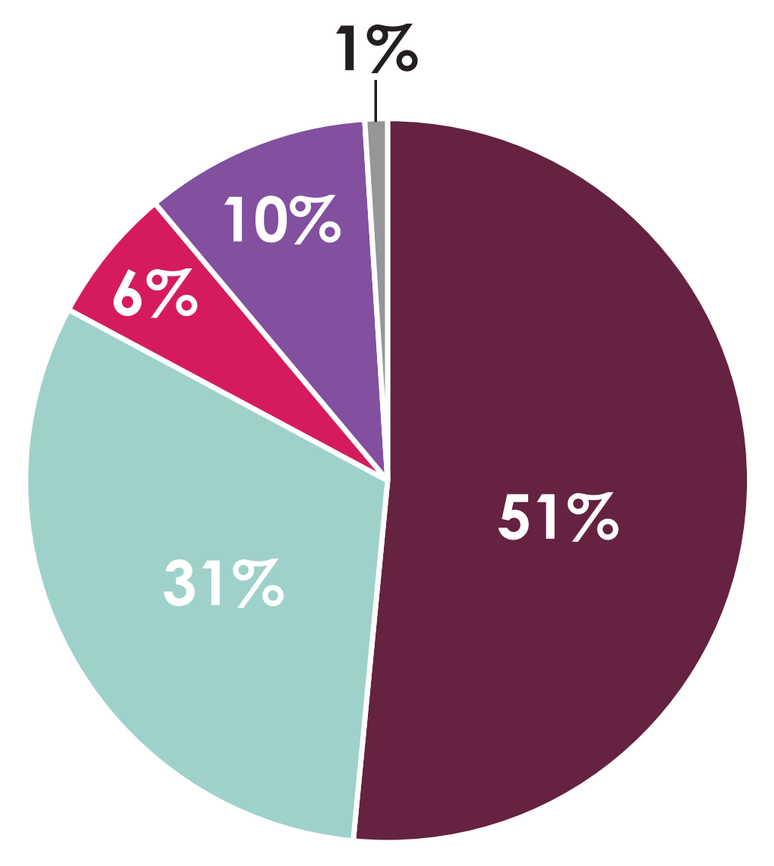

1%
10%
6%
31%
51%

	France (n=465)	Germany (n=400)	Italy (n=360)	Spain (n=361)	UK (n=336)
Platinum-based chemotherapy—cisplatin	46%	64%	48%	50%	48%
Platinum-based chemotherapy—carboplatin	35%	18%	36%	35%	29%
Other chemotherapy	8%	5%	9%	4%	2%
ICI treatment	6%	12%	6%	11%	19%
Other treatment	3%	0%	<1%	1%	0%

2L mUC treatment (n=1,777)

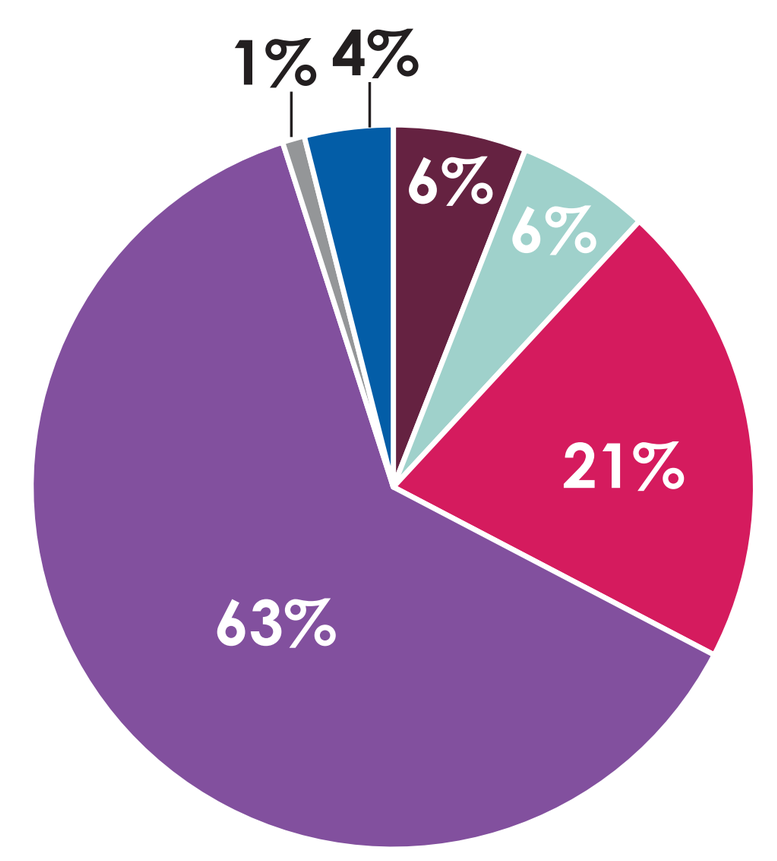

1%
4%
6%
6%
21%
63%

	France (n=268)	Germany (N=252)	Italy (n=226)	Spain (n=223)	UK (n=208)
Platinum-based chemotherapy—cisplatin	4%	9%	4%	5%	6%
Platinum-based chemotherapy—carboplatin	7%	5%	4%	7%	10%
Other chemotherapy	19%	17%	29%	22%	17%
ICI treatment	66%	63%	59%	65%	64%
Other treatment	1%	<1%	<1%	0%	0%
Best supportive care only	3%	6%	4%	2%	3%

3L mUC treatment (n=627)

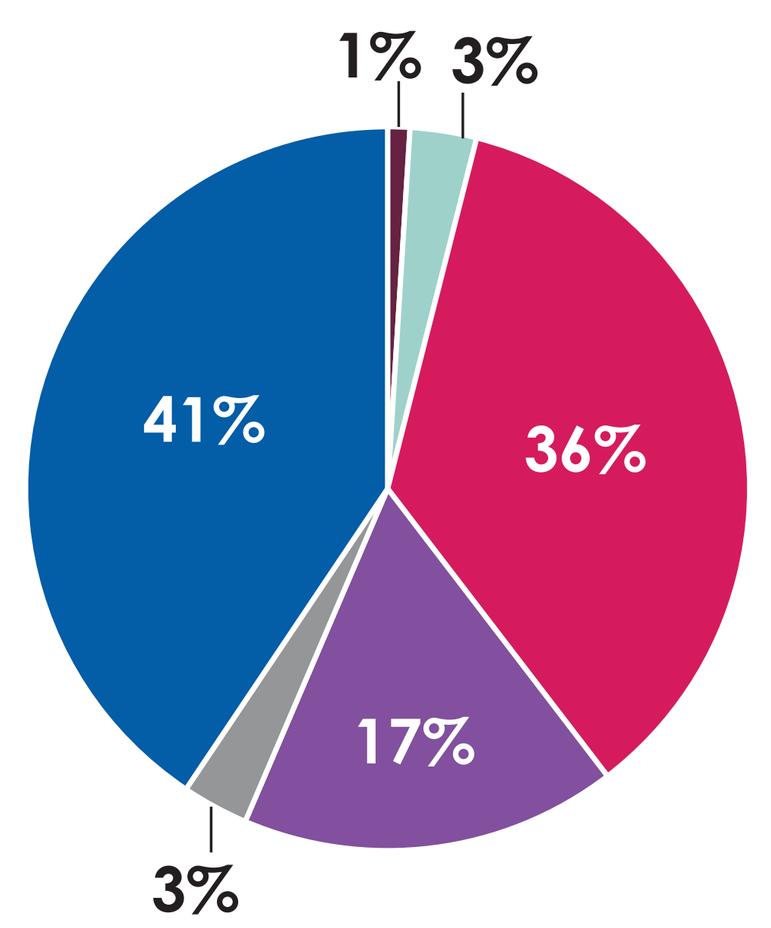

1%
3%
41%
36%
17%
3%

	France (n=140)	Germany (n=131)	Italy (n=128)	Spain (n=116)	UK (n=112)
Platinum-based chemotherapy—cisplatin	2%	2%	0%	1%	3%
Platinum-based chemotherapy—carboplatin	1%	5%	4%	0%	4%
Other chemotherapy	31%	24%	41%	53%	31%
ICI treatment	24%	20%	15%	23%	1%
Other treatment	6%	2%	0%	0%	4%
Best supportive care only	35%	48%	40%	23%	57%

- Platinum-based chemotherapy—cisplatin
- Platnium-based chemotherapy—carboplatin
- Other chemotherapy
- ICI treatment
- Other treatment
- Best supportive care only

Other chemotherapy: gemcitabine, methotrexate, vinblastine, doxorubicin, paclitaxel, docetaxel, cyclophosphamide, mitomycin C, fluorouracil, pemetrexed, ifosfamide, epirubicin, etoposide, vinflunine, and did not receive a platinum-based chemotherapy.
ICI treatment: atezolizumab, nivolumab, durvalumab, pembrolizumab, or avelumab.
Other treatment: sunitinib, axitinib, fluorouracil, niraparib, everolimus, filgrastim, oxaliplatin, enfortumab vedotin, cabozantinib, tisotumab vedotin, pazopanib, or cetrelimab.
1L, first line; **2L**, second-line; **3L**, third-line; **Eu5**, France, Germany, Italy, Spain, and UK; **ICI**, immune checkpoint inhibitor; **mUC**, metastatic urothelial cancer.

REFERENCES 1. Witjes JA, et al. *EAU Guidelines*, 2022. ISBN 978-94-92671-16-5. **2.** Powles T, et al. *Ann Oncol*. 2022;33(3):244-58. **3.** Bavencio (avelumab). Summary of product characteristics. Merck Europe B.V., Amsterdam, Netherlands, an affiliate of Merck KGaA, Darmstadt, Germany; 2022. **4.** Sung H, et al. *CA Cancer J Clin*. 2021;71:209-49. **5.** Anderson P, et al. *Curr Med Res Opin*. 2008;24(11):3063-72.

DISCLOSURES N. Milloy is an employee of Adelphi Real World. **M. Kirker** is an employee of Pfizer. **M. Berry** is an employee of Adelphi Real World. **R. Montgomery** is an employee of Adelphi, is an immediate family member of AstraZeneca, and holds stock and other ownership interests for AstraZeneca. **C. Bleasdale** is an employee of Adelphi Real World. **M. Kearney** is an employee of the healthcare business of Merck KGaA, Darmstadt, Germany. **N. Costa** is an employee, holds stock and other ownership, and has received honoraria from Pfizer. **J. Chang** is an employee of Pfizer and holds stock and other ownership interests from Bayer, Bristol Myers Squibb, and Pfizer. **ACKNOWLEDGMENTS** Adelphi Real World has received institutional research funding from Pfizer, as part of an alliance between the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945) and Pfizer.

Poster No. RWD50. Presented at ISPOR 2022; May 15-18, 2022; Washington, DC; Hybrid.