Comparison of efficacy and safety of brigatinib in first-line treatments for patients with anaplastic lymphoma kinase-positive non-small cell lung cancer: A systematic review and indirect treatment comparison

Yongfeng Yu[1], Fanfan Zhu[2], Wenxin Zhang[2] , Shun Lu[1,*]

[1]Shanghai Lung Cancer Center, Shanghai Chest Hospital, Shanghai Jiao Tong University, Shanghai, China; [2]Takeda (China) International Trading Co., Ltd, Shanghai; *Correspondence: shun_lu@hotmail.com

Background

- Drug resistance develops after crizotinib's initial benefits, particularly in the central nervous system (CNS)[1-2].
- Several next-generation ALK inhibitors, including ceritinib, alectinib, brigatinib, ensartinib and lorlatinib have been developed.
- Our objective was to evaluate the relative efficacy and safety of brigatinib compared with other ALK inhibitors for the first-line treatment of patients with *ALK*-positive non-small cell lung cancer (NSCLC).

Methods

Search strategy and selection criteria

- Electronic databases were systematically searched from January 2010 to October 2021. Studies were considered if they met the inclusion criteria as follows: (1) ALK inhibitor-naïve *ALK*-positive NSCLC patients; (2) either ALK inhibitors or chemotherapy were included in the control arms; (3) Phase III RCTs with PFS, OS, ORR and safety profile reported.
- Outcomes evaluated by indirect treatment comparison (ITC) using Bucher method.

Data extraction and quality assessment

- Data were extracted by two independent investigators, and discrepancies were resolved by involving a third investigator. When more than one article reported the same outcome, the most recent data were selected.
- The qualities of the studies were evaluated following the *Cochrane Handbook Bias Risk Assessment of Randomized Controlled Studies*[3].

Results

Studies included in the ITC

- 9 RCTs[2,4-13] (ALTA-1L, ALEX, J-ALEX, ALESIA, PROFILE1014, PROFILE 1029, ASCEND-4, eXalt3 and CROWN) with 2,484 patients assessing crizotinib, ceritinib, alectinib, brigatinib, ensartinib, and lorlatinib were included.

Figure 1: The network plot of included studies

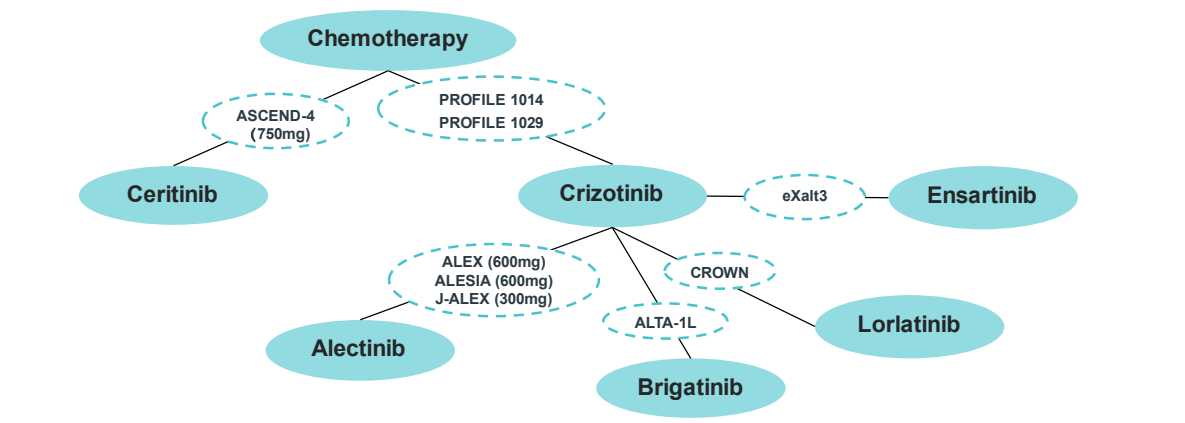
Chemotherapy
ASCEND-4 (750mg)
PROFILE 1014
PROFILE 1029
Ceritinib
Crizotinib
eXalt3
Ensartinib
ALEX (600mg)
ALESIA (600mg)
J-ALEX (300mg)
CROWN
ALTA-1L
Lorlatinib
Alectinib
Brigatinib

Risk of bias

- All studies properly reported randomized sequences generation and were at low risk for selection bias. Open-label studies were considered at high risk of bias in performance.

ITC results of efficacy endpoints

- In ITT patients, brigatinib significantly prolonged BIRC-assessed PFS compared with crizotinib and ceritinib; and had a comparable PFS with other 2nd generation ALK inhibitors. Similar PFS benefit were observed in the subgroups of Asian patients and patients with baseline brain metastases.
- In addition, the ITC results also showed numerically higher PFS with brigatinib compared with low-dose alectinib and ensartinib in patients with baseline brain metastases, though the difference was not statistically significant.

Figure 2a: PFS with brigatinib vs other ALK inhibitors in ITT patients

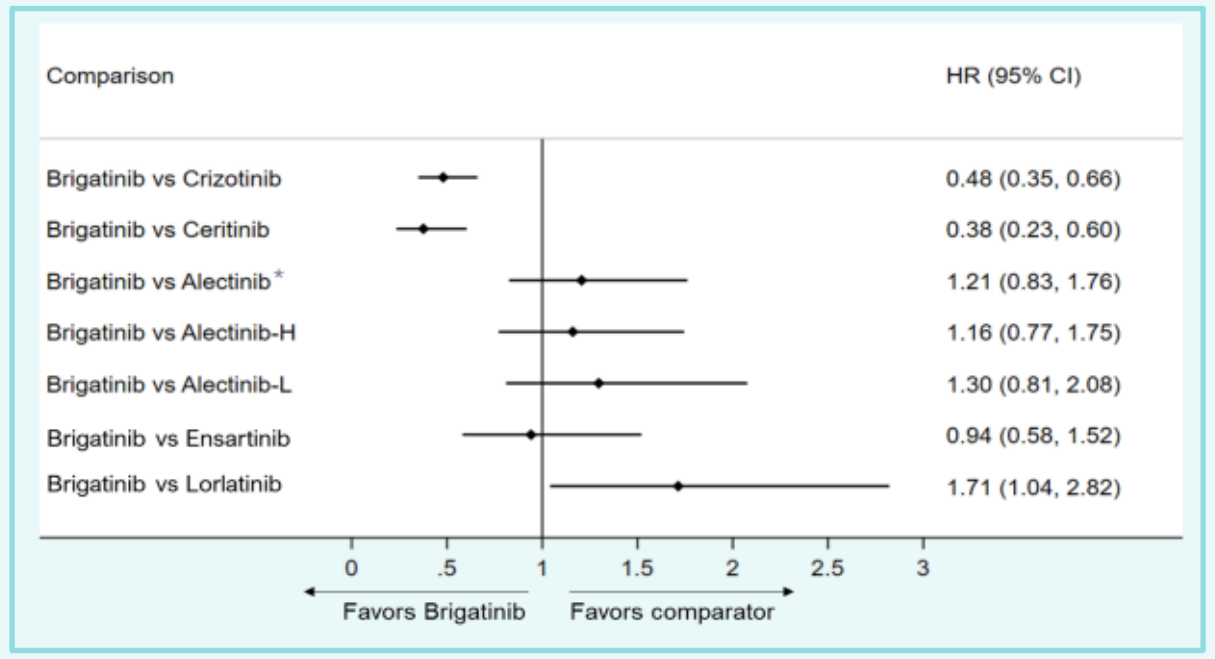
Comparison
HR (95% CI)
Brigatinib vs Crizotinib 0.48 (0.35, 0.66)
Brigatinib vs Ceritinib 0.38 (0.23, 0.60)
Brigatinib vs Alectinib* 1.21 (0.83, 1.76)
Brigatinib vs Alectinib-H 1.16 (0.77, 1.75)
Brigatinib vs Alectinib-L 1.30 (0.81, 2.08)
Brigatinib vs Ensartinib 0.94 (0.58, 1.52)
Brigatinib vs Lorlatinib 1.71 (1.04, 2.82)
0 .5 1 1.5 2 2.5 3
Favors Brigatinib
Favors comparator

*The relative benefit of alectinib versus crizotinib was demonstrated by pooled results of ALEX, ALESIA and J-ALEX; Alectinib-H (high-dose alectinib): pooled ALEX and ALESIA study results; Alectinib-L (low-dose alectinib): included J-ALEX study results

Figure 2b: PFS with brigatinib vs other ALK inhibitors in Asian patients

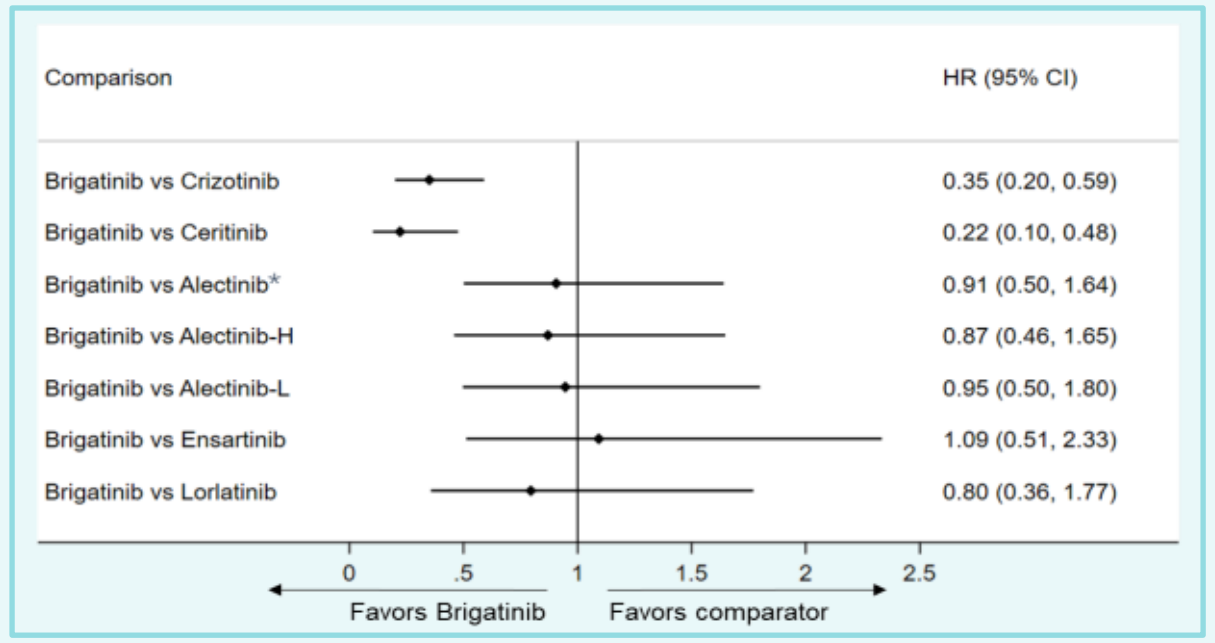
Comparison
HR (95% CI)
Brigatinib vs Crizotinib 0.35 (0.20, 0.59)
Brigatinib vs Ceritinib 0.22 (0.10, 0.48)
Brigatinib vs Alectinib* 0.91 (0.50, 1.64)
Brigatinib vs Alectinib-H 0.87 (0.46, 1.65)
Brigatinib vs Alectinib-L 0.95 (0.50, 1.80)
Brigatinib vs Ensartinib 1.09 (0.51, 2.33)
Brigatinib vs Lorlatinib 0.80 (0.36, 1.77)
0 .5 1 1.5 2 2.5
Favors Brigatinib
Favors comparator

*The relative benefit of alectinib versus crizotinib was demonstrated by pooled results of ALEX, ALESIA and J-ALEX; Alectinib-H (high-dose alectinib): pooled ALEX and ALESIA study results; Alectinib-L (low-dose alectinib): included J-ALEX study results

Figure 2c: PFS with brigatinib vs other ALK inhibitors in patients with baseline brain metastases

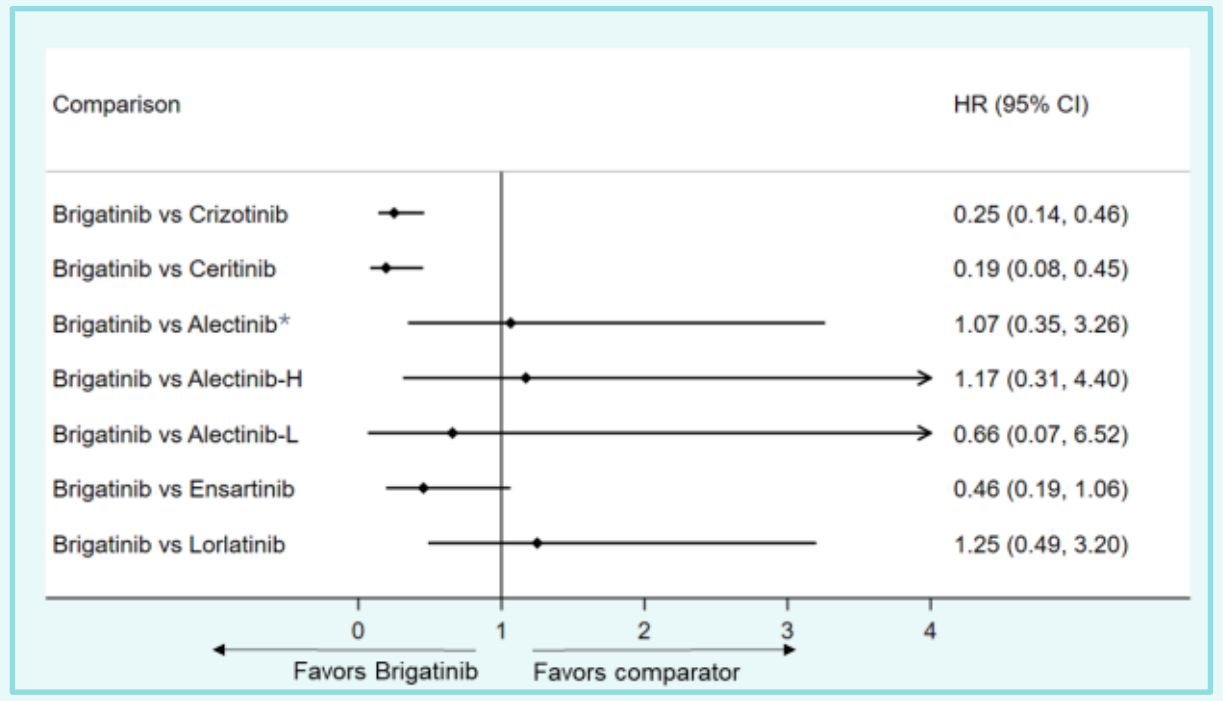
Comparison
HR (95% CI)
Brigatinib vs Crizotinib 0.25 (0.14, 0.46)
Brigatinib vs Ceritinib 0.19 (0.08, 0.45)
Brigatinib vs Alectinib* 1.07 (0.35, 3.26)
Brigatinib vs Alectinib-H 1.17 (0.31, 4.40)
Brigatinib vs Alectinib-L 0.66 (0.07, 6.52)
Brigatinib vs Ensartinib 0.46 (0.19, 1.06)
Brigatinib vs Lorlatinib 1.25 (0.49, 3.20)
0 1 2 3 4
Favors Brigatinib
Favors comparator

*The relative benefit of alectinib versus crizotinib was demonstrated by pooled results of ALEX, ALESIA and J-ALEX; Alectinib-H (high-dose alectinib): pooled ALEX and ALESIA study results; Alectinib-L (low-dose alectinib): included J-ALEX study results

- Brigatinib significantly reduced the risk of death compared with crizotinib after adjusting for treatment crossover in crizotinib arm.
- No significant differences were observed in OS between brigatinib and other next generation ALK inhibitors.

Figure 3: OS with brigatinib vs other ALK inhibitors in ITT patients

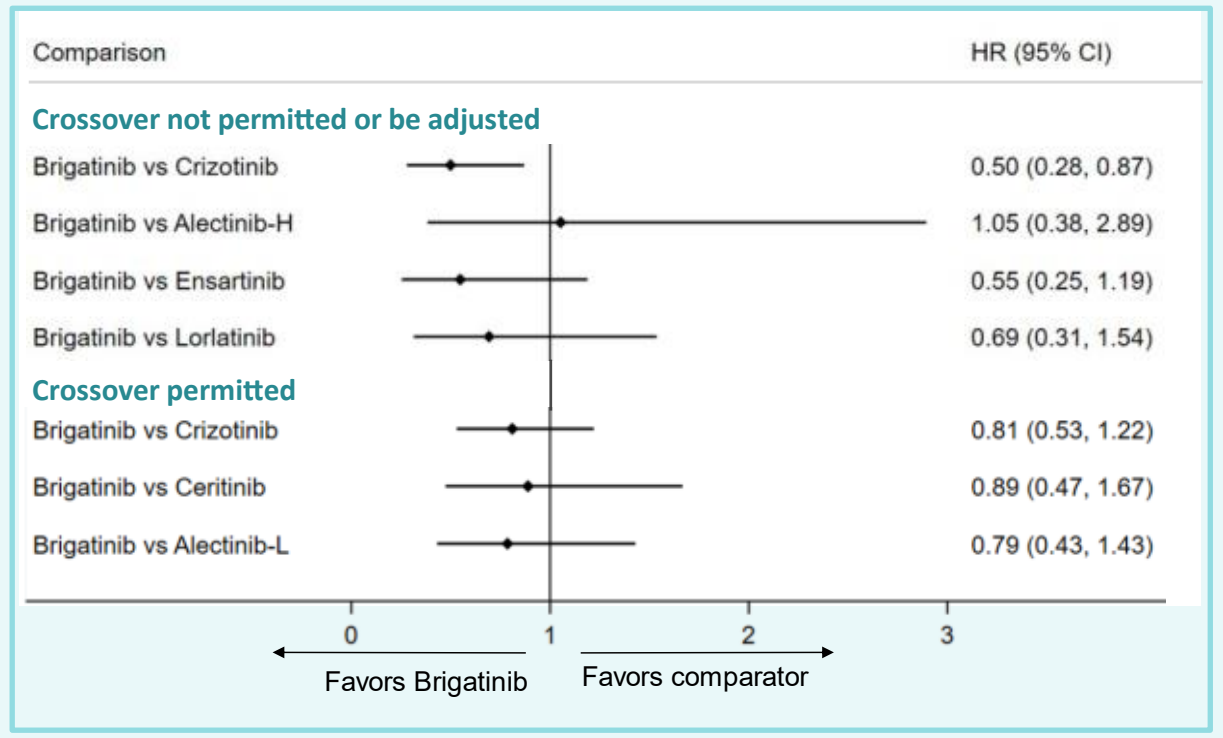
Comparison
HR (95% CI)
Crossover not permitted or be adjusted
Brigatinib vs Crizotinib 0.50 (0.28, 0.87)
Brigatinib vs Alectinib-H 1.05 (0.38, 2.89)
Brigatinib vs Ensartinib 0.55 (0.25, 1.19)
Brigatinib vs Lorlatinib 0.69 (0.31, 1.54)
Crossover permitted
Brigatinib vs Crizotinib 0.81 (0.53, 1.22)
Brigatinib vs Ceritinib 0.89 (0.47, 1.67)
Brigatinib vs Alectinib-L 0.79 (0.43, 1.43)
0 1 2 3
Favors Brigatinib
Favors comparator

*Alectinib-H (high-dose alectinib): pooled ALEX and ALESIA study results; Alectinib-L (low-dose alectinib): included J-ALEX study results

- Brigatinib was associated with better ORR than crizotinib (OR: 1.73, 95%CI: 1.04, 2.88), and comparable ORR with other ALK inhibitors.
- For patients with measurable brain metastases at baseline, brigatinib had significantly superior effects in intracranial ORR compared to crizotinib (OR: 11.67, 95%CI: 2.15, 63.27).

Figure 4: ORR with brigatinib vs other ALK inhibitors in ITT patients

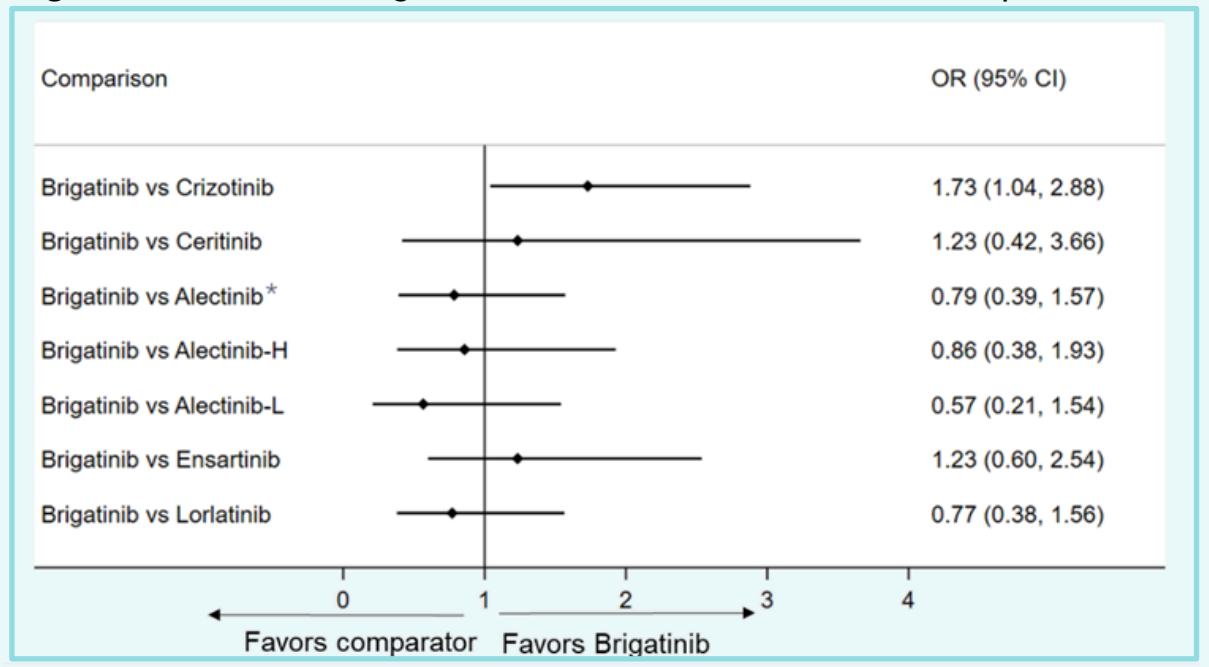
Comparison
OR (95% CI)
Brigatinib vs Crizotinib 1.73 (1.04, 2.88)
Brigatinib vs Ceritinib 1.23 (0.42, 3.66)
Brigatinib vs Alectinib* 0.79 (0.39, 1.57)
Brigatinib vs Alectinib-H 0.86 (0.38, 1.93)
Brigatinib vs Alectinib-L 0.57 (0.21, 1.54)
Brigatinib vs Ensartinib 1.23 (0.60, 2.54)
Brigatinib vs Lorlatinib 0.77 (0.38, 1.56)
0 1 2 3 4
Favors comparator
Favors Brigatinib

The relative benefit of alectinib versus crizotinib was demonstrated by pooled results of ALEX, ALESIA and J-ALEX; Alectinib-H (high-dose alectinib): pooled ALEX and ALESIA study results; Alectinib-L (low-dose alectinib): included J-ALEX study results

ITC results of safety profiles

- The incidence of grade ≥3 AEs of brigatinib is comparable to next generation ALK inhibitors (except alectinib), and no significant differences were observed in the incidence of AEs leading to discontinuation between brigatinib and other ALK inhibitors.

HRQoL endpoints

- Brigatinib significantly delayed time to worsening in the EORTC QLQ-C30 GHS/QoL vs. crizotinib (HR: 0.69, 95%CI: 0.49, 0.98) and significantly delayed time to worsening of emotional and social functioning and symptoms of fatigue, nausea and vomiting, appetite loss, and constipation (log-rank $p < 0.05$)[12].
- Due to the lack of patient level data, no further ITC analyses of HRQoL between brigatinib and other ALK inhibitors have been performed.

Discussion

- We have used BIRC-assessed and most updated trial data and considering treatment crossover.
- The relative efficacy was further analyzed in the subgroups of Asian patients and patients with baseline brain metastases.
- Intracranial efficacy was demonstrated for brigatinib in ALTA-1L[12], given the lack of a similar demonstration for other 2nd generation ALK inhibitors, no further ITC was conducted. Future studies are expected to supplement this data gap.

Conclusions

- Brigatinib was superior to crizotinib and ceritinib in PFS and had comparable efficacy and safety profile with other 2nd generation ALK inhibitors in first-line treatments for patients with *ALK*-positive NSCLC.

References

1. Dagogo-Jack, I. Ann Oncol 2016
2. Solomon, B. J, et al. N Engl J Med 2014
3. Higgins, J. P, et al. Bmj 2011,
4. Soria, J. C et al. Lancet 2017
5. Hida, T et al. Lancet 2017
6. Zhou, C et al. Lancet Respir Med 2019
7. Mok, T et al. Ann Oncol 2020
8. Selvaggi, G et al. Journal of Thoracic Oncology 2021
9. Shaw, A. T et al. N Engl J Med 2020
10. Camidge, D. R et al. N Engl J Med 2018
11. Camidge, D. R et al. J Clin Oncol 2020
12. Camidge, D. R et al. J Thorac Oncol 2021
13. Wu, Y. L et al. J Thorac Oncol 2018

Acknowledgments

Medical writing was provided by Shuli Qu and Yuxia Wu from Real World Solutions, IQVIA, Shanghai, China and funded by Takeda (China) International Trading Co., Ltd.

Funding

This study was funded by Takeda (China) International Trading Co., Ltd.

Abbreviations

- RCT: Randomized Clinical trial; ITT: Intent-to-treat
- PFS: Progression-free survival; OS, Overall survival; ORR, Objective response rate
- HRQoL: health-related quality of life; GHS: global health status;
- EORTC QLQ-C30: European Organization for Research and Treatment of Cancer Quality of Life Questionnaire-Core 30;
- BIRC: blinded independent review committee

Disclosures

Dr. Shun Lu received research support and speaker fees from AstraZeneca, Hutchison, BMS, Heng Rui, Beigene, Roche, and Hansoh, and acted as advisor and consultant with AstraZeneca, Pfizer, Boehringer Ingelheim, Hutchison MediPharma, Simcere, ZaiLab, GenomiCare, Yuhan Corporation, PrIME Oncology, Menarini, and Roche. Fanfan Zhu and Wenxin Zhang are employees of Takeda. Dr. Yongfeng Yu has nothing to disclose.

Content intended for a HCP audience only