PCR179

Development of a Conceptual Model of the Patient Experience in Small-Cell Lung Cancer (SCLC): A Qualitative Interview Study

Danielle Altman[1], An-Chen Fu[2], Patrick Marquis[1], Alissa Rams[1], Jessica Baldasaro[1], Samir Ali Ahmad[1], Michael Schlichting[3], Xinke Zhang*[2]

[1]Modus Outcomes, a Division of THREAD, Boston, MA, USA

[2]EMD Serono, Billerica, MA, USA

[3]the healthcare business of Merck KGaA, Darmstadt, Germany

*presenting author

GET POSTER PDF

CONCLUSIONS

In this study, we addressed a gap in the literature by specifically interviewing patients with SCLC, instead of lung cancer more broadly, and creating a conceptual model of the SCLC patient experience focused on clinical treatment benefit

To our knowledge, this is the first qualitative study of the lung cancer patient experience in which the sample was comprised only of patients with SCLC

Participants reported a breadth of symptoms and impacts of SCLC that affect their quality of life (QoL)

The conceptual model created for this study can be used to support the selection of questionnaires and items for an SCLC-bespoke patient-reported outcome (PRO) instrument that is aligned with current FDA guidance on core disease-related symptoms and physical and role functioning concepts to measure as PROs in oncology trials[4]

INTRODUCTION

- SCLC represents nearly 15% of all lung cancers. SCLC occurs primarily in smokers and is a more aggressive type of lung cancer[1]
- PROs are important tools in clinical management and drug development as diseases and treatments may impact patients' lives in ways that are only known by the patients
- While PRO questionnaires exist for lung cancer, none of these are SCLC-specific
- To inform PRO assessments in clinical trials of patients with SCLC and under the FDA patient-focused drug development guidance, it is vital to understand SCLC symptoms and the impacts of SCLC on QoL from the perspective of patients with SCLC[2]
- However, the patient experience of SCLC is not well documented in qualitative lung cancer literature, with most studies reflecting the lung cancer experience more broadly or the non-small cell lung cancer experience specifically
- It is important to fill this gap in the literature by collecting information directly from patients with SCLC on the symptoms and daily life impacts they experience

OBJECTIVES

This study aimed to identify symptoms and impacts important to patients with SCLC in the United States (US) and to develop a conceptual model that describes their experience, with a focus on concepts that relate to clinical treatment benefit

METHODS

Figure 1. Inclusion criteria

18 years or older	Diagnosed with SCLC by their doctor (self-report)	Received at least 1 line of systemic treatment (self-report)	Willing and able to participate in a 60-minute telephone or online interview	Able to read and speak English

Figure 2. Study methods

Qualitative concept elicitation interviews
- Institutional Review Board approval
- Purposive sample of participants in the US
- Semi-structured interview guide, using a primarily inductive approach[3]*
- One-on-one interviews by telephone or online
- Audio recordings transcribed and de-identified

Coding and analysis
- Team-based coding and conceptual and thematic analysis using ATLAS.ti[3]
- Saturation analysis conducted to confirm the appropriate sample size**

Conceptual model development
- Concepts (codes) were grouped based on FDA guidance on core **disease-related symptoms and physical and role functioning concepts** to measure as PROs in oncology trials[4]
- Other QoL concepts excluded, due to focus on clinical treatment benefit
- Treatment experience concepts excluded, due to treatment variation across study sample

*Patients were first asked to describe SCLC symptoms and impacts on daily life at length and in their own words. Following this, a deductive approach was used to ask patients if they had experienced other major symptoms or impacts identified in lung cancer literature

**Saturation was evaluated at the subdomain level by ordering interviews chronologically, then grouping interviews into one group of six transcripts and an additional four groups of five transcripts. Concepts emerging from the second group were compared with the concepts that emerged in the first group. The comparison was repeated for each additional group until minimal or no new information emerged

RESULTS

- Conceptual saturation was achieved in the fourth group of transcripts assessed
- The conceptual model reflects the following concepts identified in this study:
 - Disease-related symptoms, including symptoms that affect the lungs (organ-specific symptoms) and symptoms that affect the whole body (systemic symptoms)
 - Impacts of SCLC on daily life, including physical or role-functioning concepts and two standalone impacts (difficulty sleeping at night and weight loss) (**Figure 3**)

Table 1. Participant demographic information

Variable*	Total N=26
Age (years)	
Mean (standard deviation)	61.32 (11.50)
Sex n (%)	
Female	19 (73.1%)
Ethnicity n (%)	
Non-Hispanic/Non-Latino	21 (80.8%)
Race n (%)	
Black/African American	4 (15.4%)
Native Hawaiian/Pacific Islander	1 (3.8%)
White/Caucasian	17 (65.4%)
Education level n (%)	
High school graduate/GED equivalent	6 (23.1%)
College	4 (15.4%)
Associate degree	3 (11.5%)
Bachelor's degree	5 (19.2%)
Post-graduate degree	4 (15.4%)
Trade/technical certification	1 (3.8%)
Other	1 (3.8%)
Employment status n (%)	
Full time	3 (11.5%)
Homemaker	6 (23.1%)
Retired	8 (30.8%)
Disabled	6 (23.1%)
Not employed	1 (3.8%)
Treatments received	
Surgery	3 (11.5%)
Radiation	10 (38.5%)
Immunotherapy	2 (7.7%)
Intralesional chemotherapy	4 (15.4%)
Systemic chemotherapy	19 (73.1%)
Other	4 (15.4%)

*Missing data (n=1) included in calculation of percentages

Figure 3. Conceptual model of clinical treatment benefit in SCLC

Disease-related symptoms		Impacts
Systemic symptoms	**Organ-specific symptoms**	**Physical functioning**
Appetite loss	**Cough**	Symptoms limiting daily activities
Dizziness	Coughing up blood	Reduced mobility/ walking
Pain	Persistent cough	Unable to get out of bed
Pain limiting activities	Phlegm	Endurance
Fatigue	Hoarseness	Strength
Tiredness	**Chest pain/arm or shoulder pain**	Bending over
Feeling weak	Chest tightness	Squatting
Needing to sit/lie down	Chest pain	Balance
Lack of energy	Arm/shoulder pain	**Role functioning**
Thinking clearly/ concentration problems due to mental tiredness	**Difficulty breathing**	Stopping or limiting work or hobbies
	Difficulty breathing (in general)	**Impact on sleep**
	Difficulty breathing (at rest)	Difficulty sleeping at night
	Difficulty breathing (upon exertion)	**Weight loss**
	Wheezing	

Disclosures:
Danielle Altman, Patrick Marquis, Alissa Rams, Jessica Baldasaro, and Samir Ali Ahmad are employees of Modus Outcomes, a Division of Thread, and were contracted by EMD Serono, Billerica, MA, USA, to perform this research.
An-Chen Fu and Xinke Zhang are employees of EMD Serono and Michael Schlichting is an employee of the healthcare business of Merck KGaA, Darmstadt, Germany.

Funding and Acknowledgements:
This research was funded by EMD Serono (CrossRef Funder ID:10.13039/100004755). Editorial assistance (editing, formatting and typesetting the poster) was provided by Janeen Nuttall of Bioscript Stirling, Macclesfield, UK, and funded by the healthcare business of Merck KGaA, Darmstadt, Germany.

Presented at ISPOR 2022 | May 15–18, 2022 | Washington, DC, USA

References:
1. Byers LA, Rudin CM. *Cancer* 2015;121:664–672.
2. US Food and Drug Administration. Patient-Focused Drug Development: Methods to Identify What Is Important to Patients | FDA, 2019.
3. Miles MB, Huberman AM, Saldaña J. Qualitative data analysis: A methods sourcebook: Sage publications, 2018.
4. Core Patient-Reported Outcomes in Cancer Clinical Trials: Draft Guidance for Industry, 2021.