Cohort Optimizer

A Software Solution that Uses Real-World Data and AI to Optimize Criteria of Oncology Trials

Bhandari Sudip*[1], Mann Jeremy*[1], Hajela Ragesh[2], Govil Vibhor[2], Greatti Yves[1], Maguire Rick[1], Powers Jamie[1], Southwick Steve[1], Mueller Judith[1]

[1]ConcertAI, Cambridge, MA, USA, [2]ConcertAI, Bengaluru, India
* Co-first authors

Introduction

- Recent work demonstrates that many clinical trial criteria negatively affect enrollment without significantly improving the health of the cohort.
- We developed a software solution, Cohort Optimizer (CO) that uses Real-World Data (RWD) and AI to modify criteria to improve the size, overall survival, and diversity of the cohort.
- Unlike many proposed solutions which only eliminate criteria, CO optimizes the cohort by intelligently relaxing or tightening criteria.

Method

- CO allows the user to assign priorities to each characteristic of the cohort—size, overall survival, and diversity throughout the US.
- CO estimates characteristics using ConcertAI's RWD that includes 5.4 million individuals, gathered from oncology clinics throughout the US.
- After imputing missing patient characteristics, CO calculates the size, survival statistics, and diversity of the baseline cohort and candidate cohorts.
- To efficiently search through the high-dimensional space of criteria and arrive at a recommendation, CO uses an evolutionary algorithm and a custom fitness function.
- As validation, we implemented CO on 15 oncological clinical trials, translated into the Observational Medical Outcomes Partnership Common Data Model.
- For all trials, we prioritized total enrollment over survival and diversity, while ensuring that the overall survival of the recommended cohort be no less than 95% of the survival of the baseline cohort.

Figure 1: Implementation of Cohort Optimizer

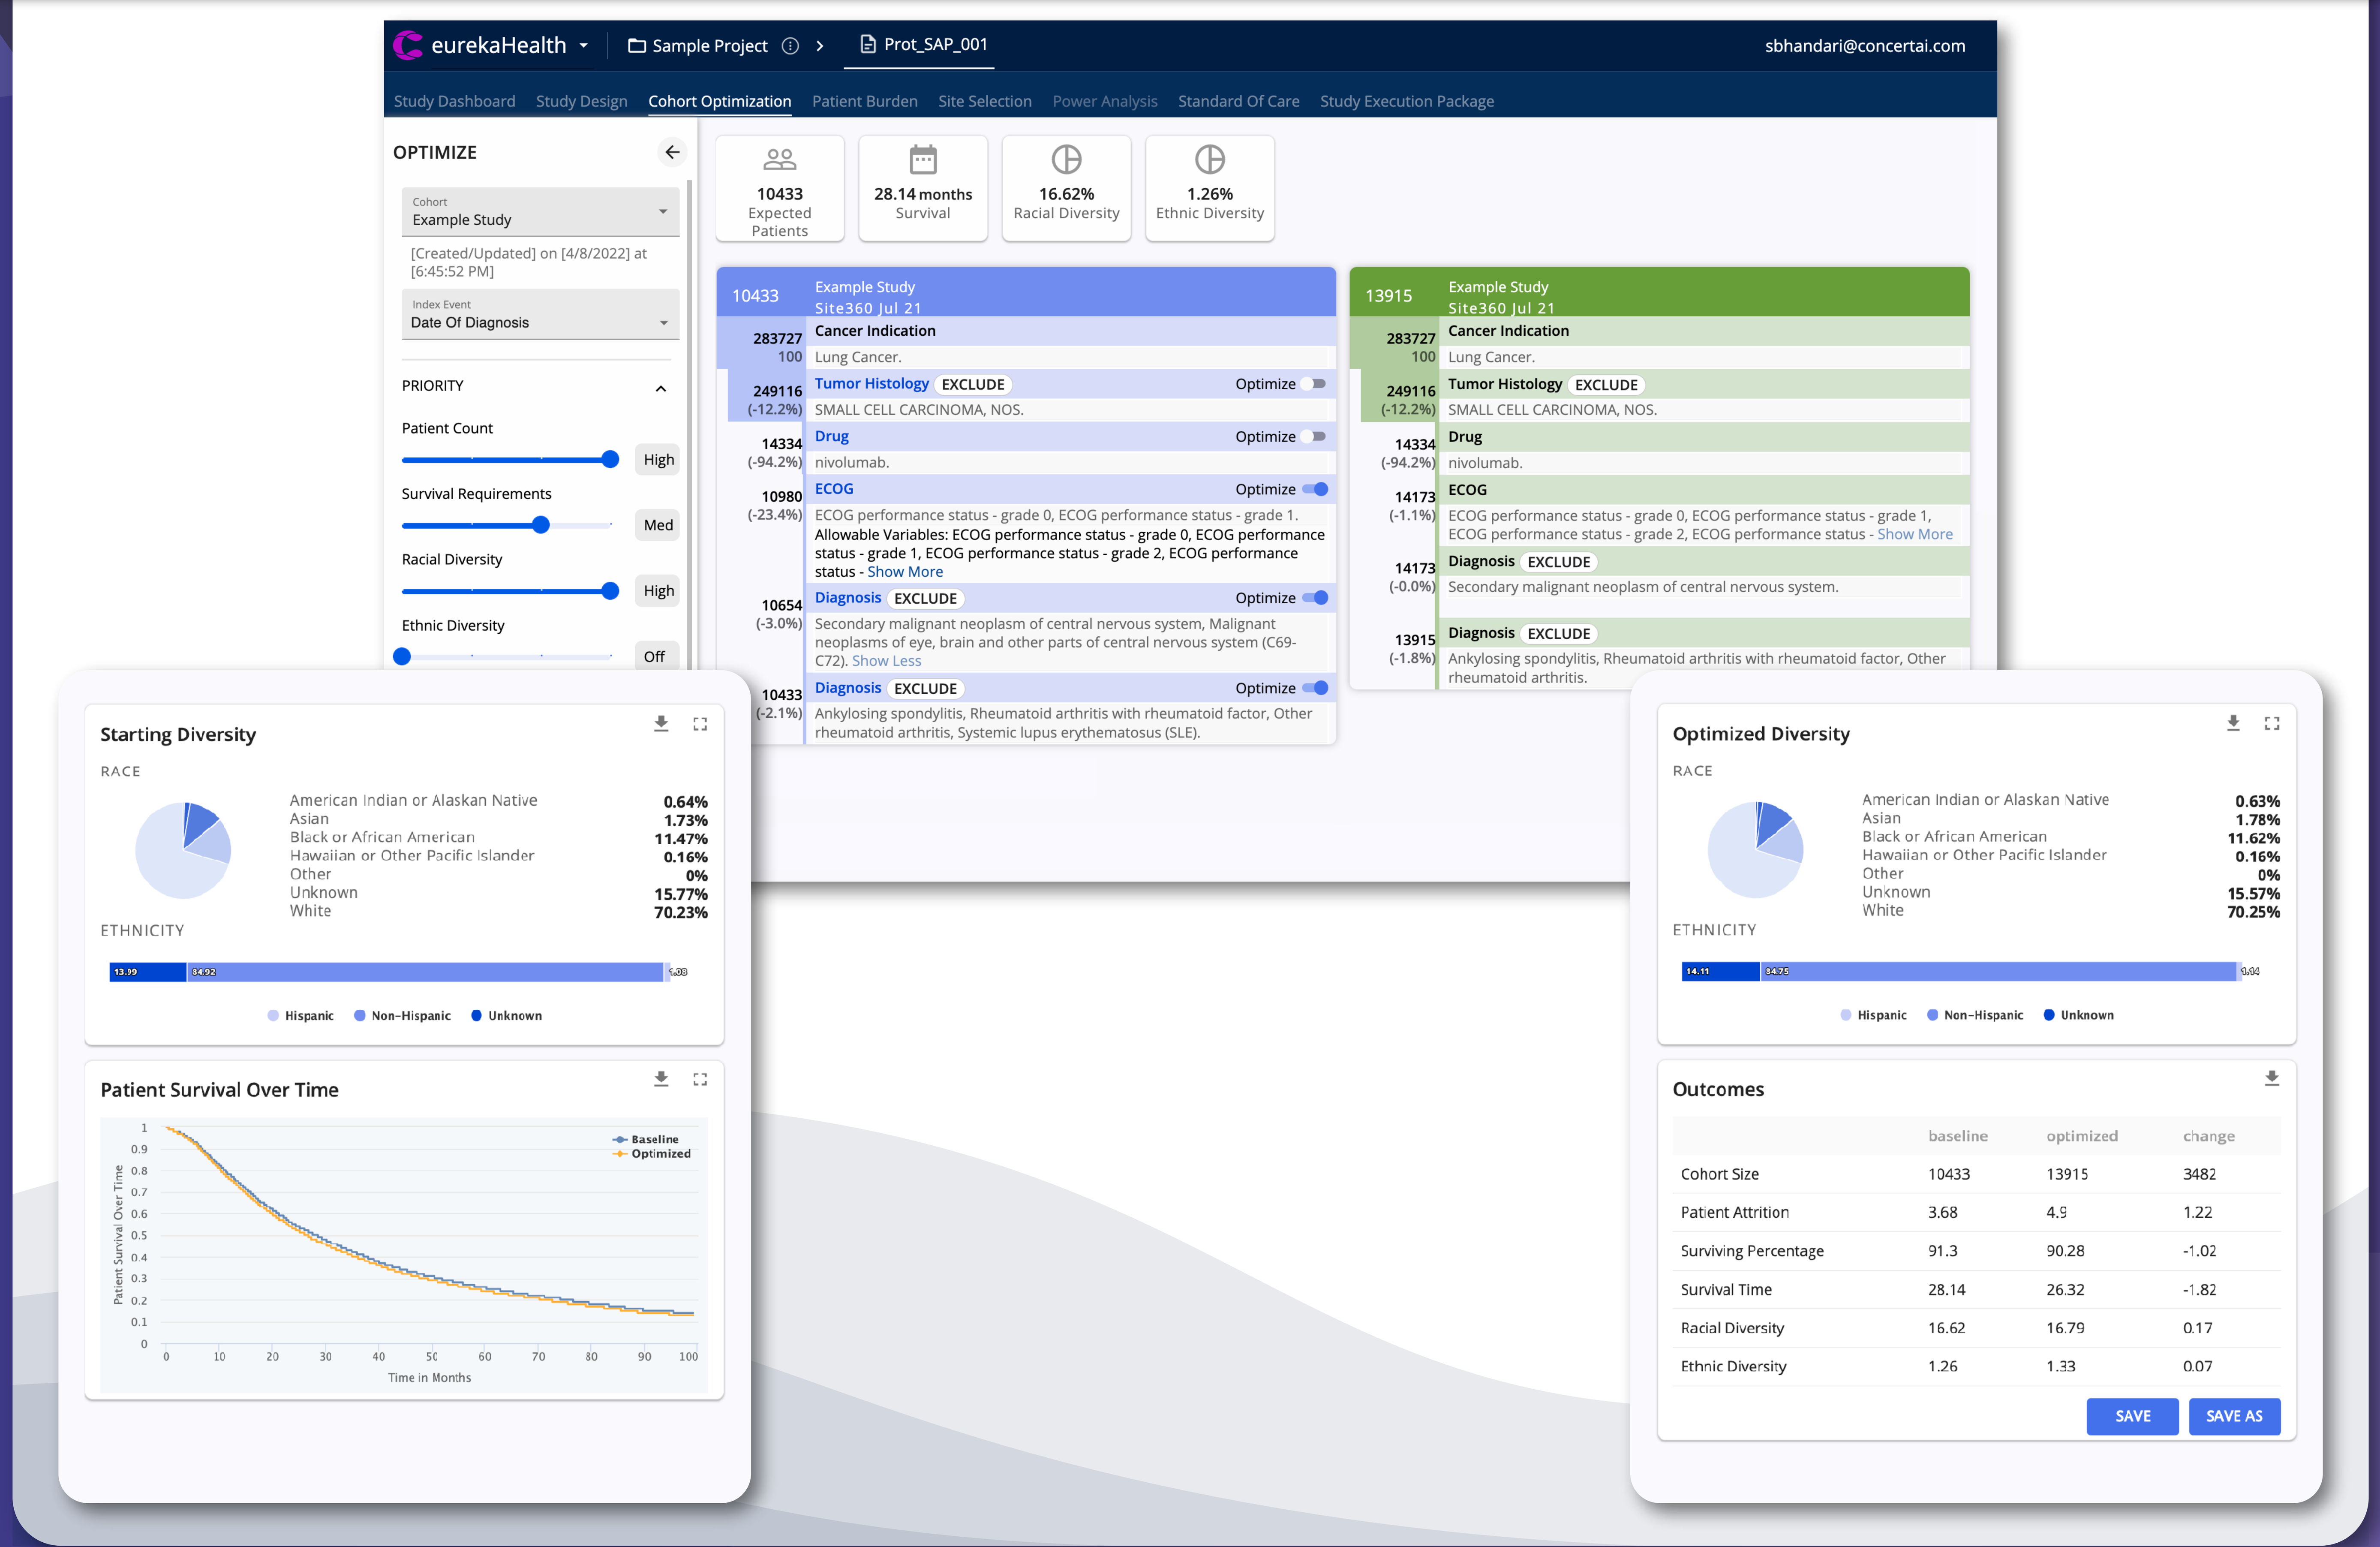
eurekaHealth
Sample Project
Prot_SAP_001
sbhandari@concertai.com
Study Dashboard
Study Design
Cohort Optimization
Patient Burden
Site Selection
Power Analysis
Standard Of Care
Study Execution Package
OPTIMIZE
Cohort
Example Study
[Created/Updated] on [4/8/2022] at [6:45:52 PM]
Index Event
Date Of Diagnosis
PRIORITY
Patient Count
High
Survival Requirements
Med
Racial Diversity
High
Ethnic Diversity
Off
10433 Expected Patients
28.14 months Survival
16.62% Racial Diversity
1.26% Ethnic Diversity
Example Study
Site360 Jul 21
Cancer Indication
Lung Cancer.
Tumor Histology EXCLUDE
SMALL CELL CARCINOMA, NOS.
Drug
nivolumab.
ECOG
Diagnosis EXCLUDE
Optimize
Starting Diversity
RACE
American Indian or Alaskan Native 0.64%
Asian 1.73%
Black or African American 11.47%
Hawaiian or Other Pacific Islander 0.16%
Other 0%
Unknown 15.77%
White 70.23%
ETHNICITY
Hispanic
Non-Hispanic
Unknown
Patient Survival Over Time
Baseline
Optimized
Patient Survival Over Time
Time in Months
Optimized Diversity
American Indian or Alaskan Native 0.63%
Asian 1.78%
Black or African American 11.62%
Hawaiian or Other Pacific Islander 0.16%
Other 0%
Unknown 15.57%
White 70.25%
Outcomes
baseline optimized change
Cohort Size 10433 13915 3482
Patient Attrition 3.68 4.9 1.22
Surviving Percentage 91.3 90.28 -1.02
Survival Time 28.14 26.32 -1.82
Racial Diversity 16.62 16.79 0.17
Ethnic Diversity 1.26 1.33 0.07
SAVE
SAVE AS

Results

- On average, our modifications increased patient counts by 75.64% compared to the baseline, without significant sacrifices in either survival or diversity.
- Much of the changes in these cohorts were attributable to relaxation (not outright elimination) of the ECOG criterion.

Conclusions

- Our method indicates that one can increase patient enrollment without compromising the overall health of the cohort.
- We demonstrate the value of RWD in estimating how changes to criteria affect the characteristics of its associated cohort. Given representative data, we believe our solution can be applied in clinical trials beyond oncology.

Contact

Sudip Bhandari, PhD
sbhandari@concertai.com

Jamie Powers, DrPH
jpowers@concertai.com

References

1. Liu X, Shi C, Deore U, et al. A Scalable AI Approach for Clinical Trial Cohort Optimization. ArXiv210902808 Cs Q-Bio. Published online September 6, 2021. Accessed October 21, 2021. http://arxiv.org/abs/2109.02808
2. Jain NM, Culley A, Knoop T, Micheel C, Osterman T, Levy M. Conceptual Framework to Support Clinical Trial Optimization and End-to-End Enrollment Workflow. JCO Clin Cancer Inform. 2019;(3):1-10. doi:10.1200/CCI.19.00033
3. Liu R, Rizzo S, Whipple S, et al. Evaluating eligibility criteria of oncology trials using real-world data and AI. Nature. Published online April 7, 2021:1-5. doi:10.1038/s41586-021-03430-5n.