The effect of lock-down on reducing hospitalisations associated with COVID-19 by States in Australia

Seulki Choi[1], Hansoo Kim[2]
[1]Sanofi, Australia*
[2]Griffith University, Australia

Griffith UNIVERSITY
Centre for Applied Health Economics

* The findings, opinions and assertions contained in this poster are those of the author. They do not necessarily represent the official positions of Sanofi.

Background

The COVID-19 response by the Australian government was focused on limiting the influx of infectious cases from overseas and flattening the curve by imposing public health measures to limit the spread. This was relatively successful in containing new cases to minimal numbers.

It was the State and Territory governments with which the primary responsibility lies for managing the impact of COVID-19. Their responsibility included investigating cases, tracing contacts and imposing restrictions. A more stringent form of universal mobility restriction commonly called “lock-down” was also a part of their responsibility.

Without a vaccine, it became evident during the first wave that lock-down was practically the only effective measure to contain the spread. Each State and Territory government had different approaches on imposing lock-downs. This is expected to have resulted in different outcomes between States and Territories.

In this analysis, the three largest States according to population, New South Wales (NSW), Victoria (VIC), and Queensland (QLD), will be investigated.

Objectives

The objective of this research was to assess the effect of lock-down on reducing COVID-19 hospitalisations and compare the effect of the lockdowns between the three largest States in Australia.

Methods

Data for COVID-19 cases, hospitalisations, vaccination roll-out and lockdown was obtained from State government websites. A analytic model was constructed to test the relationship between lock-down and hospitalisation. Lock-down and state were main predictor variables with hospitalisation being the dependant variable. Other predictor variables likely to influence the effect included active cases and vaccination. Analyses were performed in Excel and SAS.

Results

Differences between States were observed for cases and hospitalisations. Examples of a wave in each State are presented in the charts.

For NSW and VIC, the cases presented are similar. Initially a lock-down was imposed for five or seven days respectively and then was extended further.

However, other than the first wave, the QLD lock-downs were snap lock-downs lasting three to five days followed by a potential extension.

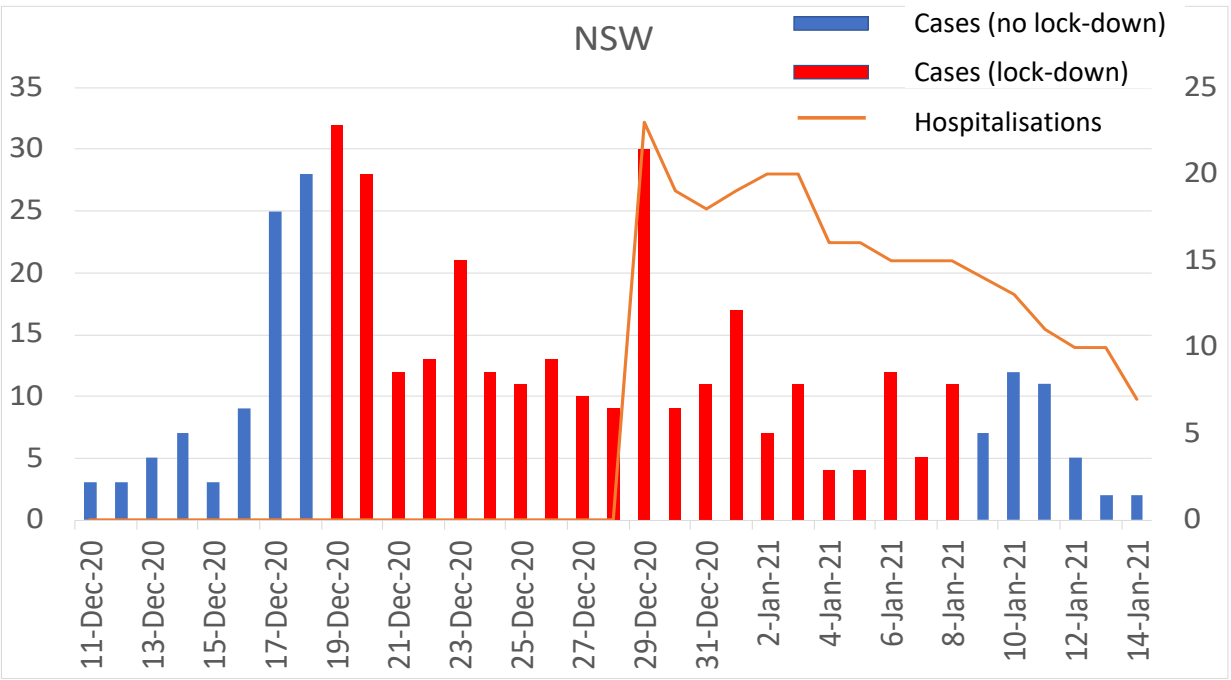
NSW
Cases (no lock-down)
Cases (lock-down)
Hospitalisations

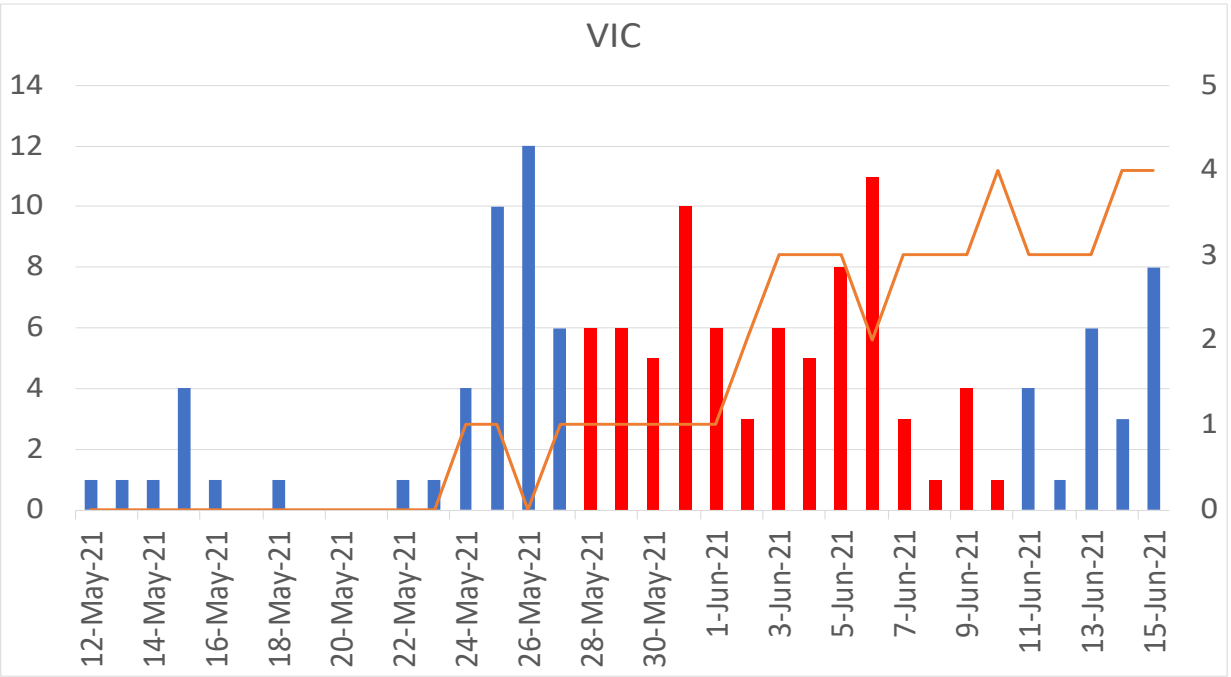
VIC

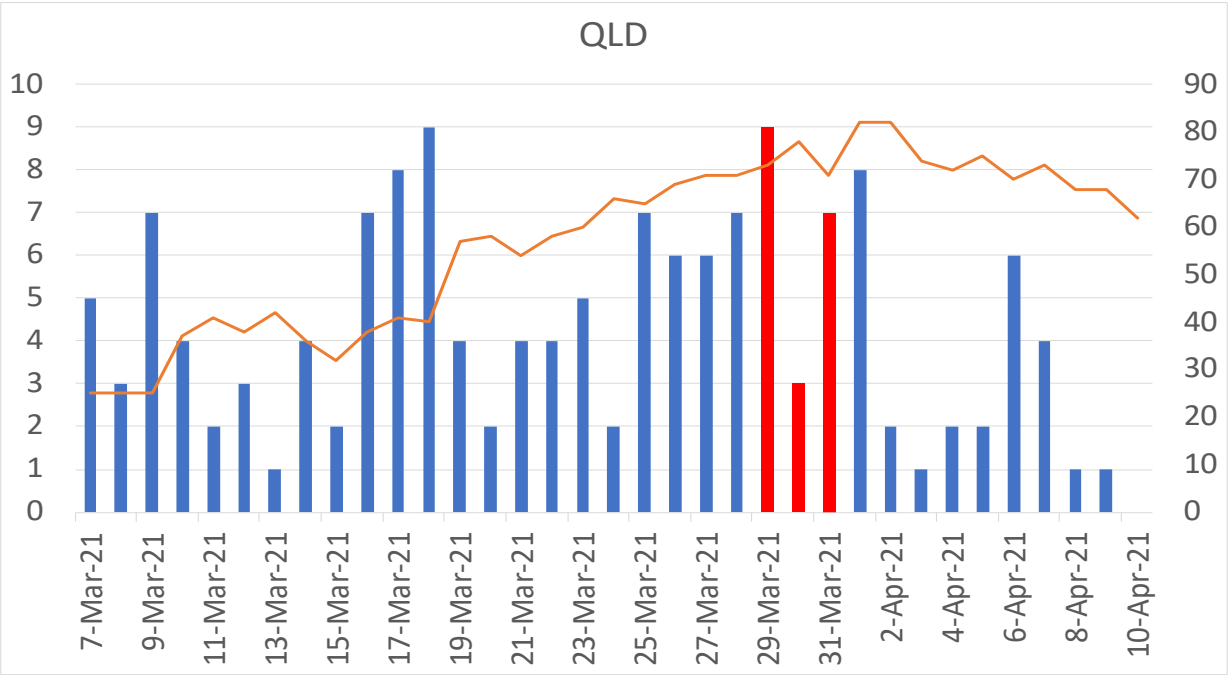
QLD

Overall, the analysis found that hospitalisation was significantly associated with lock-down ($p<0.0001$). It was observed that the total days of lock-down and the number of hospitalisations varied considerably between States.

States	Lockdown Total = 596 days (%)	Hospitalisations Mean (SD)
NSW	174 (29.2)	131.53 (301.23)
VIC	273 (45.8)	95.15 (198.82)
QLD	54 (9.1)	16.98 (16.89)

QLD had the lowest hospitalisations (M=16.98, SD=16.89) and shortest lock-down days (54 out of 596), confirming that QLD was least affected by COVID-19.

VIC had the highest number of lock-down days (273 out of 596) with the second highest number of hospitalisations (M=95.15, SD=198.82). Between VIC and QLD, the effect of lock-down on hospitalisations was not statistically significant.

On the other hand, when VIC is compared with NSW, where lock-down was shorter (174 out of 596) and hospitalisations were highest (M=131.53, SD=301.23), the effect of lock-down was significantly different ($p<0.0001$).

Discussions

No significance of lock-down effect between VIC and QLD could indicate that the strategy of QLD was effective and more efficient, however in this interpretation caution is required as the case hospitalisation ratio was observed to be much lower in QLD.

The effect of lock-downs have been assessed in various approaches and contexts. Our approach differs from previous analyses in that difference of lock-down implementation was assessed as a nested condition.

Conclusion

Our analysis indicates that lock-down is associated with reduction of hospitalisations. The snap lock-down strategy in QLD did not require an extension in most of lock-downs, which could indicate that the strategy was successful. VIC and QLD have largely different number of days in lock-down, however the impact of lock-down was not significantly different. This could imply that hospitalisations were effectively brought down with the lock-down strategy in VIC.