Real-World Treatment Patterns among Patients with Mycosis Fungoides and Sézary Syndrome in the United States between 2018 and 2020

Y. Zhao, PhD[1]; J. Tang, MSc[1]; H. Zhou, MSc[2]; T. Takahashi, PhD[3], T. Nomura, PhD[3], E. Dennis, MD[1]
[1]Kyowa Kirin, Inc., Bedminster, NJ, USA; [2]KMK Consulting, Morristown, NJ, USA; [3]Kyowa Kirin Co, Ltd., Tokyo, Japan

Introduction

- Cutaneous T cell lymphomas (CTCL) are a rare, heterogenous group of non-Hodgkin lymphomas with an annual incidence of 6–10 per million persons and a prevalence of approximately 40,000 persons in the United States (US) [1-3].
- Mycosis fungoides (MF) and its leukemic variant Sézary syndrome (SS) are the most common subtypes of CTCL, comprising approximately 70% of cases [4,5].
- The chronic and recurrent nature of these lymphomas frequently requires patients to receive repeated lines of treatment and maintenance therapies [6].
- Given their low prevalence, real-world treatment patterns of existing and newly approved therapies for MF and SS are not currently well-characterized, especially during the COVID pandemic.

Objectives

- This study examined treatment patterns among patients with MF or SS between 2018-2020 in the US

Methods

Data Source

- This study used de-identified patient-level data from Symphony Health Solutions' Integrated Dataverse database (SHS), a longitudinal patient data source that tracks 274 million active patients in the US
 - SHS data are integrated from medical practices, pharmacies, and hospitals to longitudinally assess healthcare delivery and patient usage patterns
 - Data are representative of the US population in terms of age, sex, geography and insurance coverage
- Medical, prescription, and hospital claims data were linked through de-identified patient codes

Population

- Patients with ≥1 diagnosis code for MF or SS (ICD-10-CM: C84.xx) in each of 2018, 2019, and 2020 were classified into 2 mutually exclusive cohorts: 1) MF only (no SS diagnosis), 2) SS

Variables

- Patient demographics (i.e., age, sex)
- All treatments recommended by the National Comprehensive Cancer Network guidelines, version 2.2021
 - Skin-directed therapy: topical, local radiation, total skin electron beam therapy, and phototherapy
 - Systemic therapy: extracorporeal photopheresis, parenteral, and oral
 - Bone marrow transplant
- Treatments were identified using national drug codes, current procedural terminology and healthcare common procedure coding system codes

Analyses

- Descriptive statistics were reported for the patient characteristics
- Treatment usage rates were reported by patient cohort

Results

- Overall, 10,527, 10,078, and 9,414 patients had ≥1 MF diagnosis (and no SS diagnosis) in 2018, 2019 and 2020 (mean age: 62.9, 63.2 and 63.2 years; male: 54.6%, 54.3% and 53.9%), respectively
- Overall, 869, 882, and 853 patients had ≥1 SS diagnosis in 2018, 2019 and 2020 (mean age: 66.3, 66.9 and 67.3 years; male: 54.4%, 54.8% and 55.6%), respectively

Treatment rates for patients with MF and SS in 2018, 2019 and 2020

	MF						SS					
	2018 (N=10,527)		2019 (N=10,078)		2020 (N=9,414)		2018 (N=869)		2019 (N=882)		2020 (N=853)	
	n	%	n	%	n	%	n	%	n	%	n	%
Any therapy	5,922	56.3	5,653	56.1	5,324	56.6	561	64.6	597	67.7	587	68.8
Systemic therapy	1,331	12.6	1,219	12.1	1,274	13.5	363	41.8	396	44.9	397	46.5
Extracorporeal photopheresis	76	0.7	57	0.6	47	0.5	148	17.0	132	15.0	115	13.5
Parenteral systemic	561	5.3	506	5.0	562	6.0	180	20.7	240	27.2	245	28.7
Oral systemic	850	8.1	783	7.8	799	8.5	155	17.8	137	15.5	151	17.7
Skin directed therapy	5,482	52.1	5,280	52.4	4,940	52.5	425	48.9	460	52.2	451	52.9
Topical	4,250	40.4	4,216	41.8	4,095	43.5	368	42.3	409	46.4	412	48.3
Local radiation	481	4.6	473	4.7	484	5.1	67	7.7	68	7.7	57	6.7
Total skin electron beam therapy	463	4.4	458	4.5	476	5.1	62	7.1	71	8.0	59	6.9
Phototherapy	1,710	16.2	1,500	14.9	1,201	12.8	55	6.3	44	5.0	35	4.1
Bone marrow transplant	12	0.1	8	0.1	20	0.2	13	1.5	12	1.4	7	0.8

Note: Patients with multiple treatments were counted in many categories

Results

Distribution by sex among patients with MF

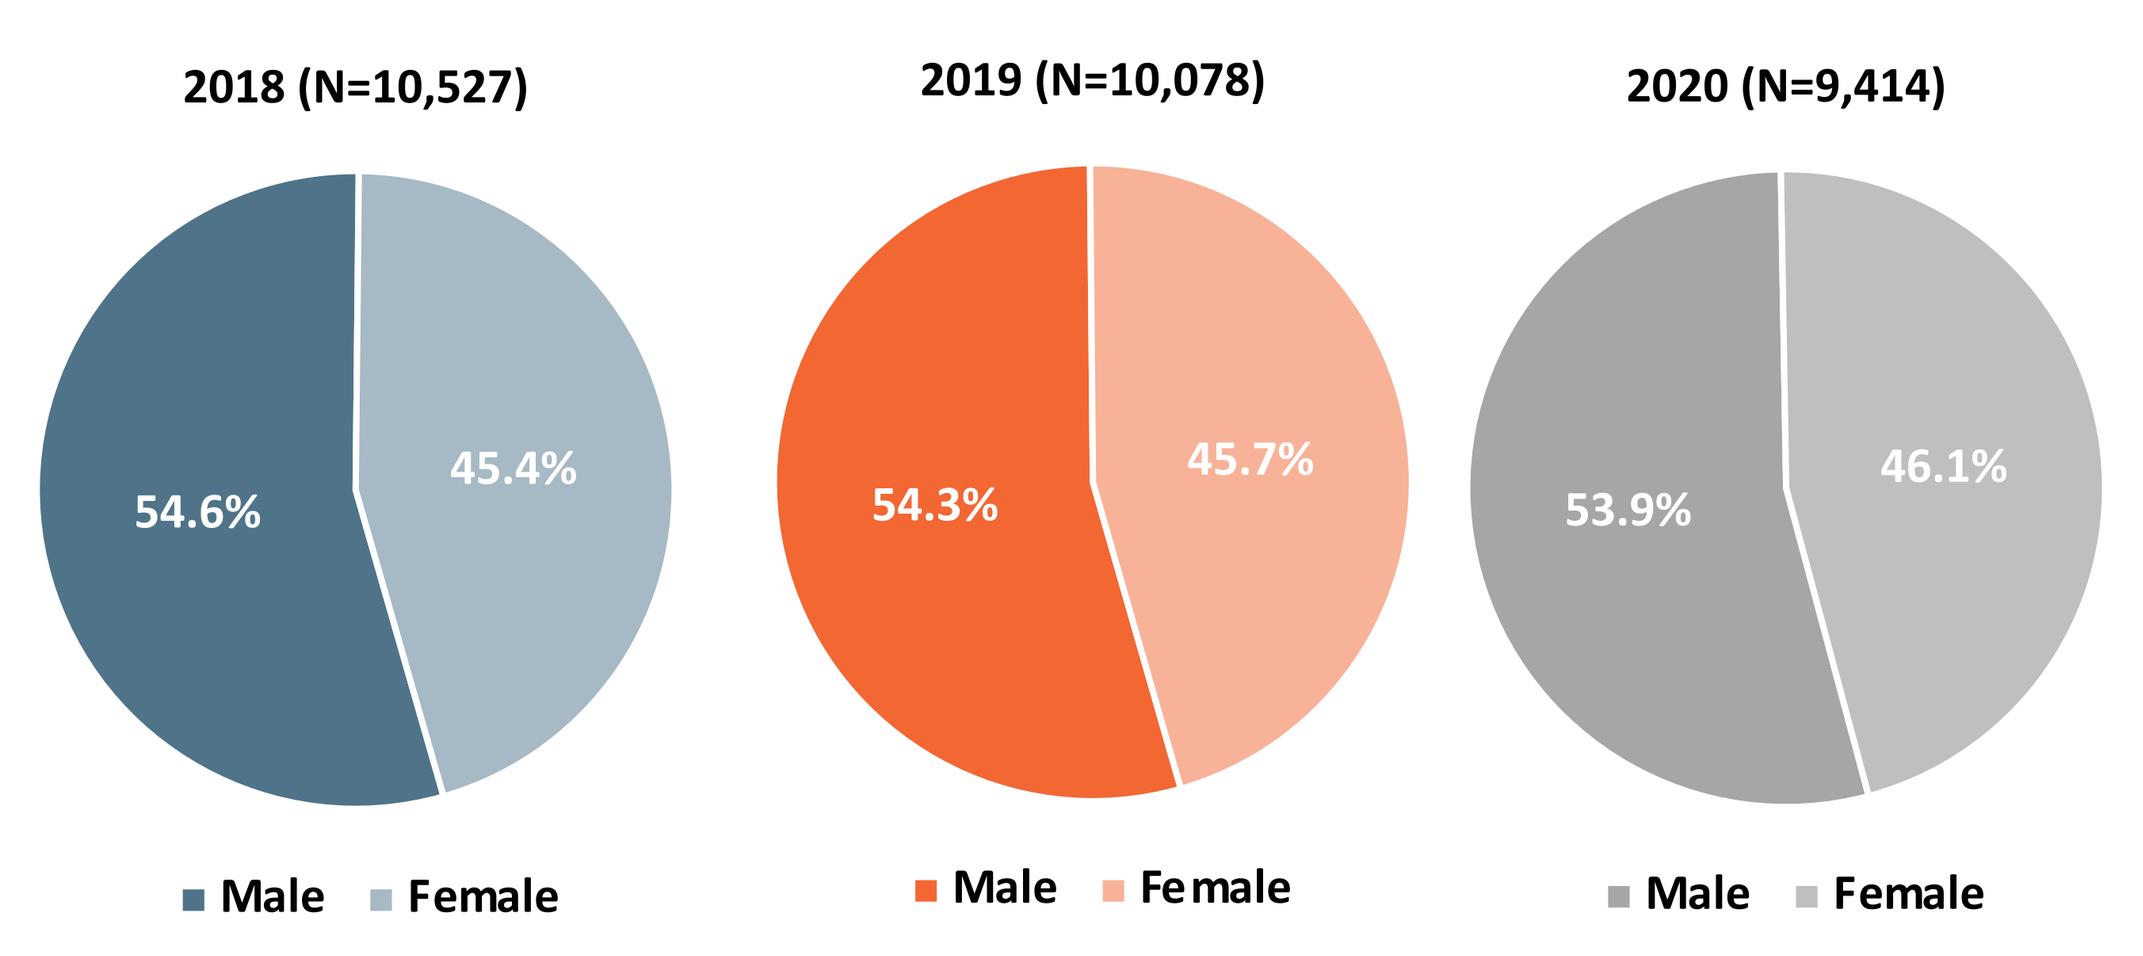
2018 (N=10,527)
54.6%
45.4%
2019 (N=10,078)
54.3%
45.7%
2020 (N=9,414)
53.9%
46.1%
Male
Female

Most commonly used systemic therapies among patients with MF

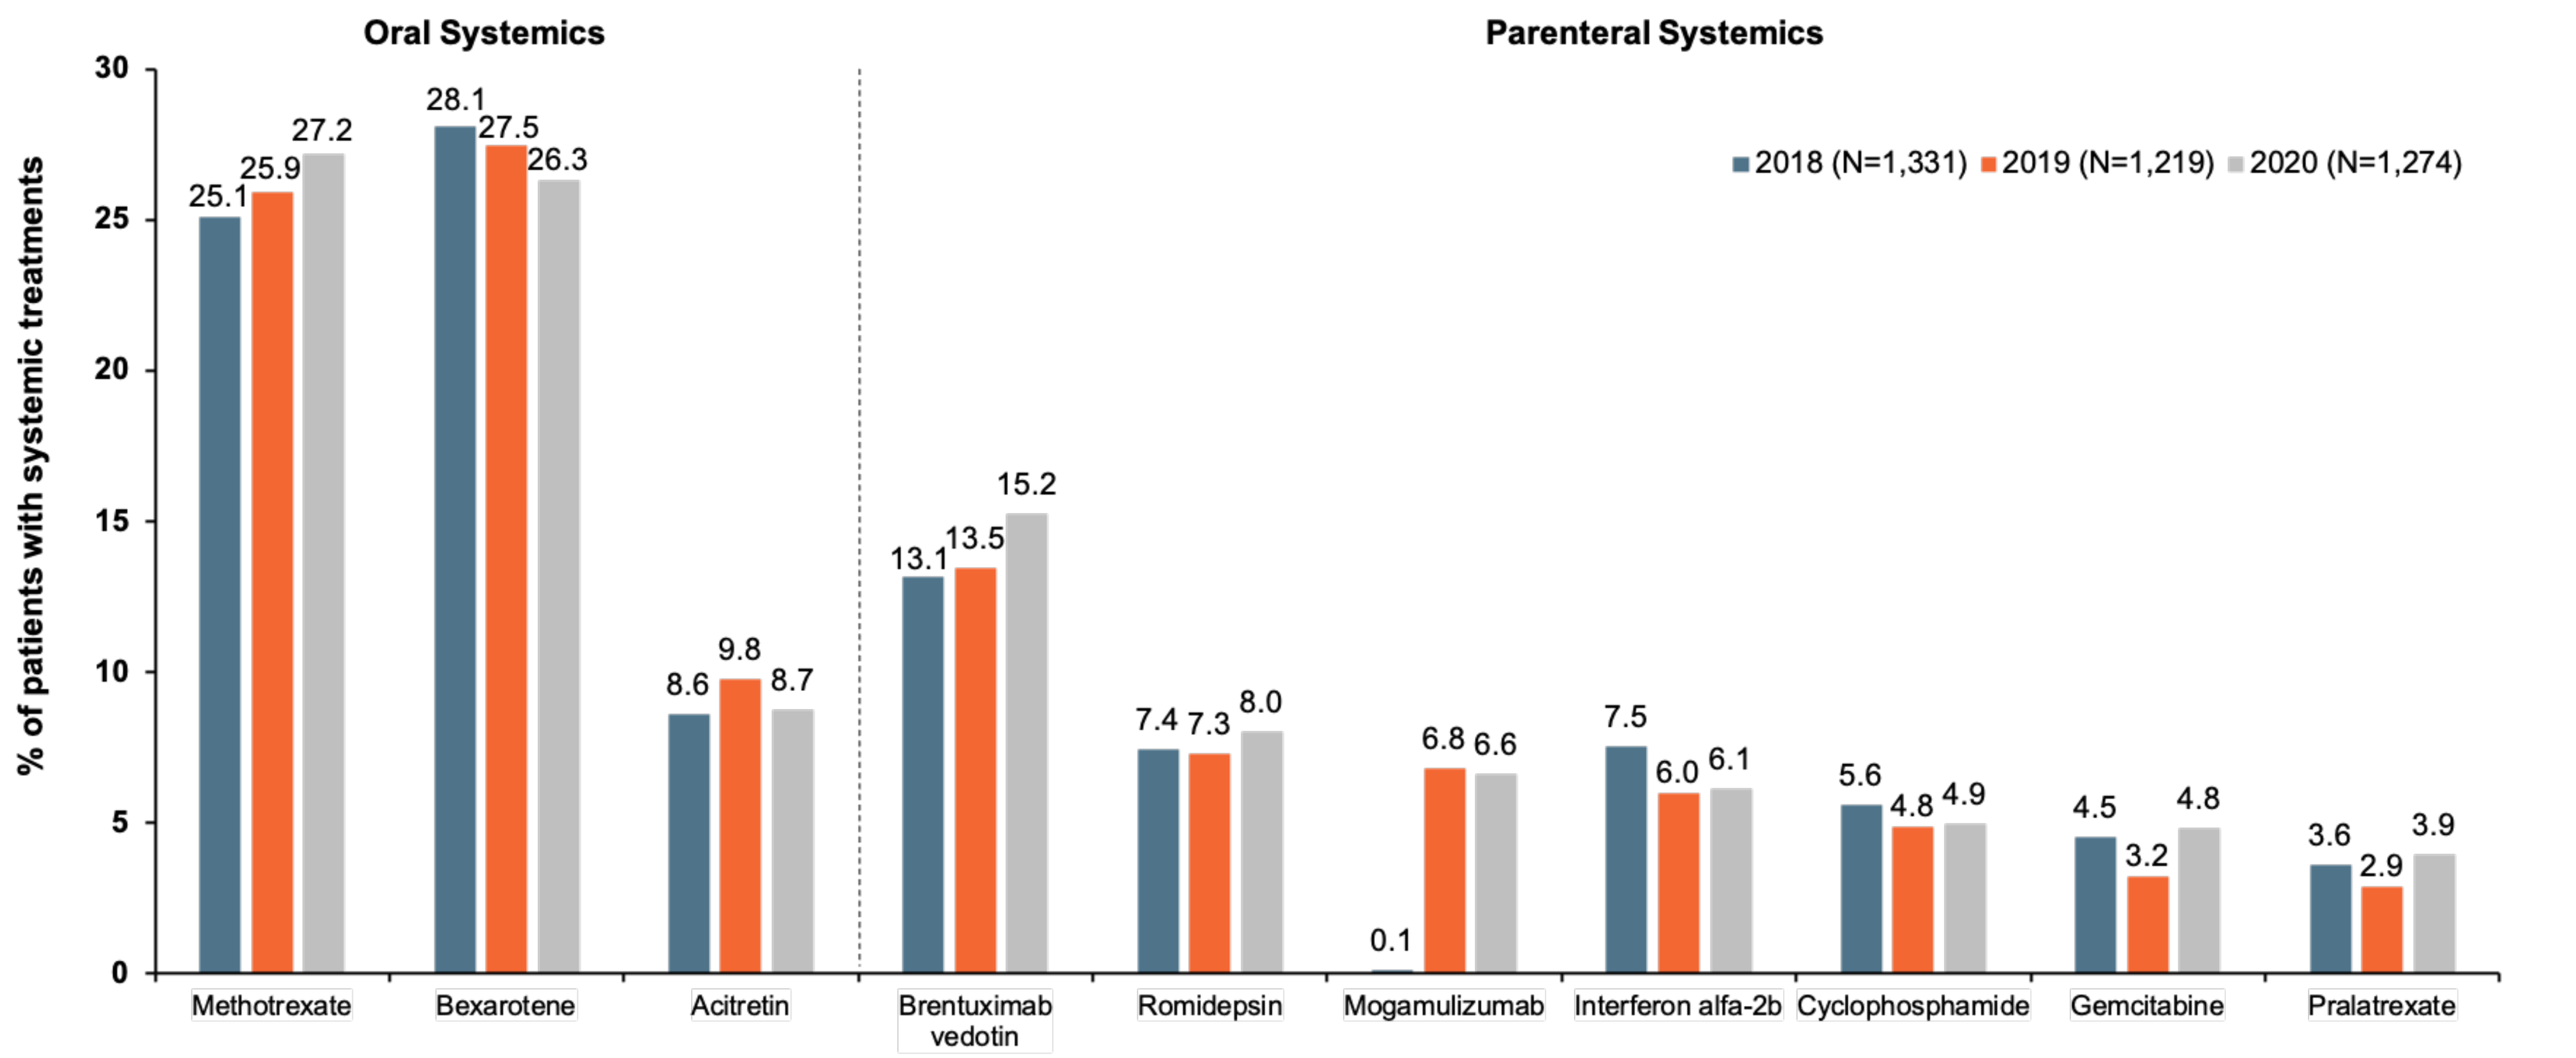
Oral Systemics
Parenteral Systemics
2018 (N=1,331) 2019 (N=1,219) 2020 (N=1,274)
% of patients with systemic treatments
Methotrexate 25.1 25.9 27.2
Bexarotene 28.1 27.5 26.3
Acitretin 8.6 9.8 8.7
Brentuximab vedotin 13.1 13.5 15.2
Romidepsin 7.4 7.3 8.0
Mogamulizumab 0.1 6.8 6.6
Interferon alfa-2b 7.5 6.0 6.1
Cyclophosphamide 5.6 4.8 4.9
Gemcitabine 4.5 3.2 4.8
Pralatrexate 3.6 2.9 3.9

Distribution by sex among patients with SS

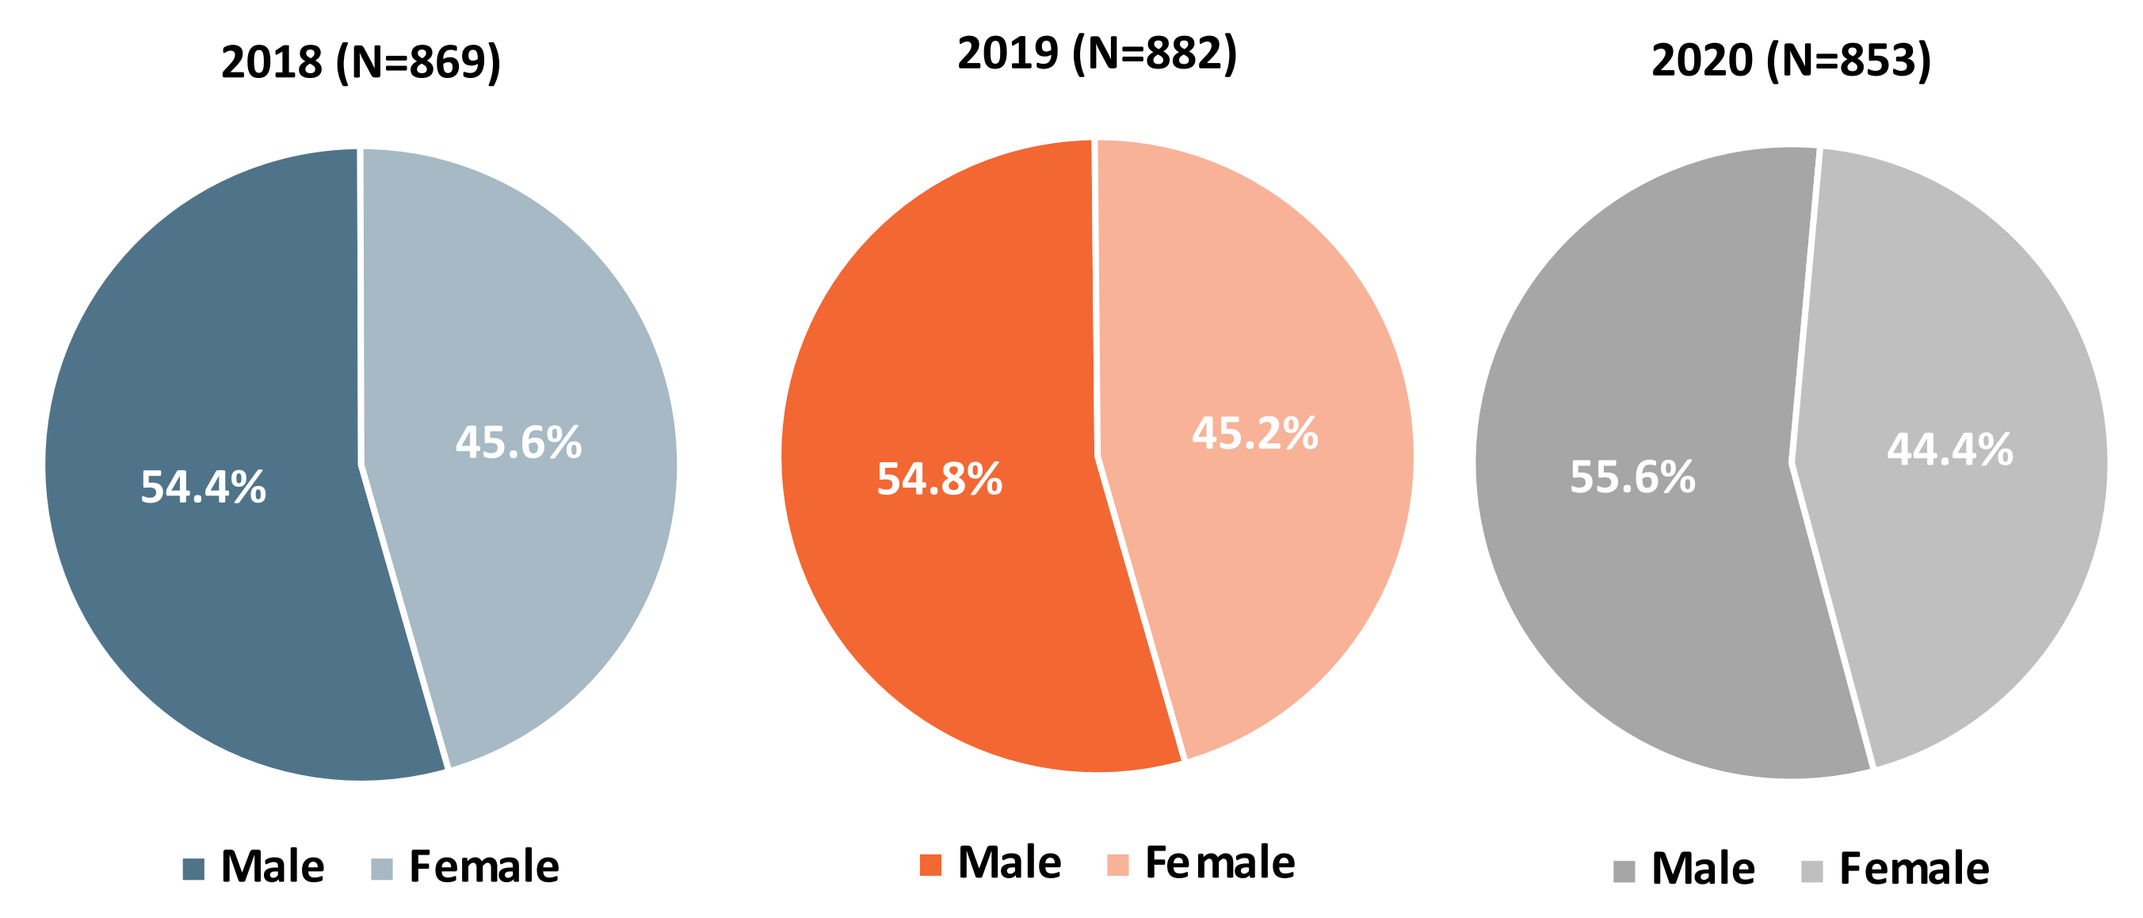
2018 (N=869)
54.4%
45.6%
2019 (N=882)
54.8%
45.2%
2020 (N=853)
55.6%
44.4%
Male
Female

Most commonly used systemic therapies among patients with SS

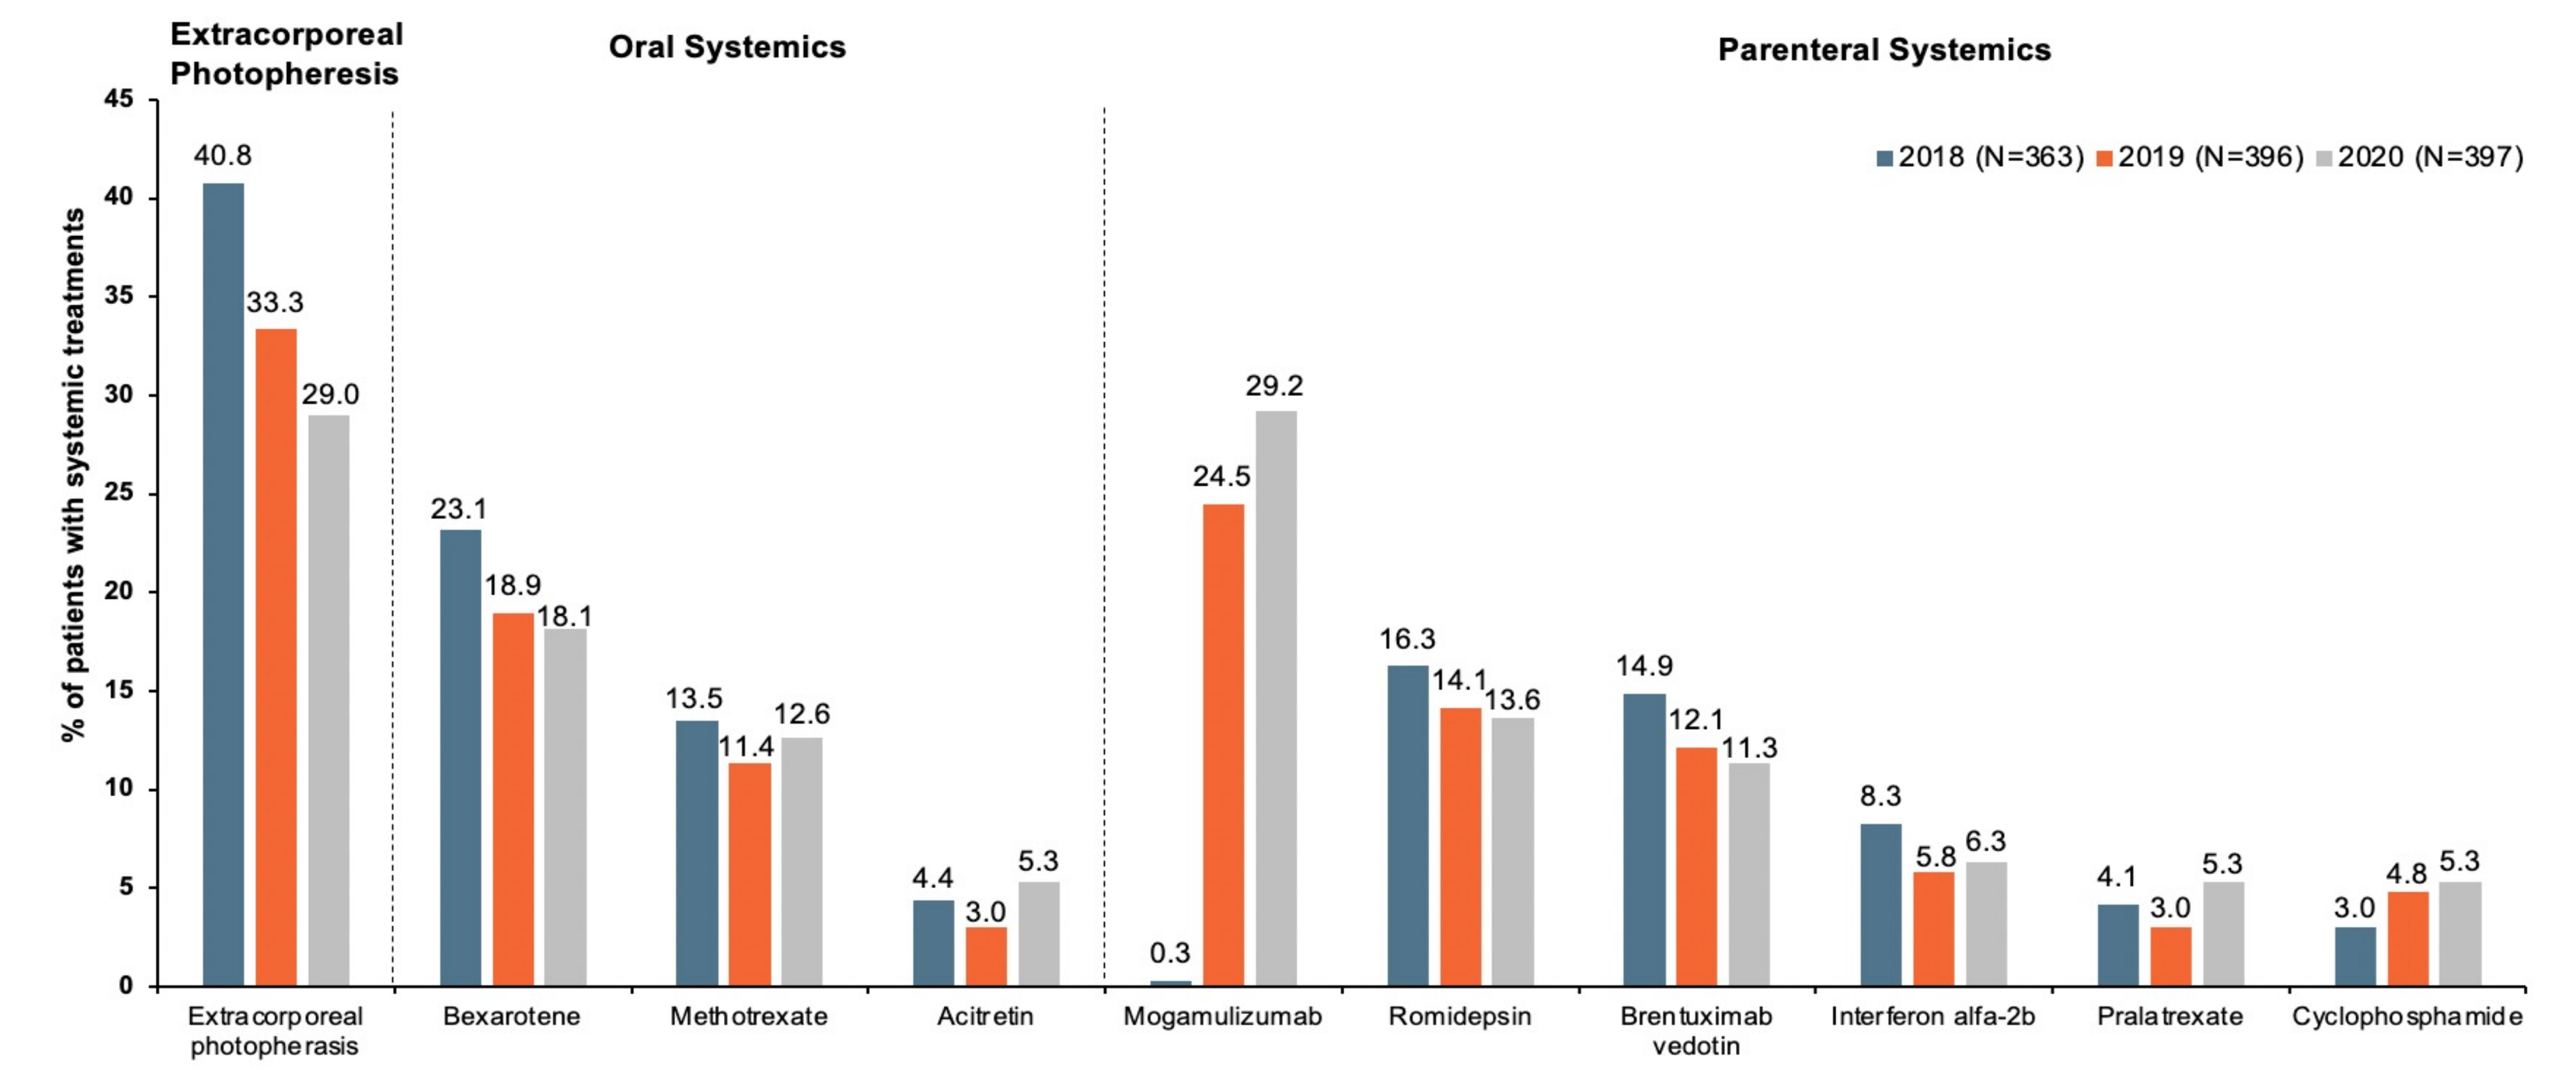
Extracorporeal Photopheresis
Oral Systemics
Parenteral Systemics
2018 (N=363) 2019 (N=396) 2020 (N=397)
% of patients with systemic treatments
Extracorporeal photopherasis 40.8 33.3 29.0
Bexarotene 23.1 18.9 18.1
Methotrexate 13.5 11.4 12.6
Acitretin 4.4 3.0 5.3
Mogamulizumab 0.3 24.5 29.2
Romidepsin 16.3 14.1 13.6
Brentuximab vedotin 14.9 12.1 11.3
Interferon alfa-2b 8.3 5.8 6.3
Pralatrexate 4.1 3.0 5.3
Cyclophosphamide 3.0 4.8 5.3

Note: All numbers were calculated among patients treated with any systemic therapies. Patients with multiple treatments were counted in many categories. The order of therapies was based on most commonly used systemic therapies in 2020.

Limitations

- Patients with MF and SS were identified using diagnosis codes, which may result in misclassification due to coding error and patients may not be identified due to incomplete diagnosis capture
- All MF or SS treatments were identified using claims captured in the SHS database, therefore may not reflect the full utilization
- Information on disease severity was not available
- Study results may not be generalizable to a population with different insurance coverage (commercial, Medicare, Medicaid) or patient characteristics (i.e., age, sex)

Conclusions

- Claims data from the 2018-2020 SHS database indicated that approximately half of MF and SS patients used skin directed therapy each year
- Among MF patients, methotrexate, bexarotene, and brentuximab were the most used systemic therapies across 3 years (each with >13% use)
 - Mogamulizumab remained at ~7% in 2019 and 2020, and was the 3rd most used among parenteral systemic therapies
- Among SS patients, mogamulizumab was the most prescribed systemic therapy in 2020, representing 29.2% use among SS patients with any systemic treatment
 - Extracorporeal photopheresis, bexarotene, romidepsin, and brentuximab had reduced utilization over time

References

1. Korgavkar K, Xiong M, Weinstock M. Changing incidence trends of cutaneous T-cell lymphoma. *JAMA Dermatol*. 2013; 149(11): 1295–1299.
2. Dummer R, et al. Cutaneous T cell lymphoma. *Nat Rev Dis Primers*. 2021; 7(1), 61.
3. Rosenberg AR, Semenov YR, Herbosa C, Mehta-Shah N, Musiek AC. Health-related quality of life and economic implications of cutaneous T-cell lymphoma. *Br J Dermatol*. 2020; 182(1), 190-196.
4. Dobos G, et al. Epidemiology of cutaneous T-cell lymphomas: A systematic review and meta-analysis of 16,953 patients. *Cancers (Basel)*. 2020; 12(10), 2921.
5. Imam MH, et al. Incidence and survival patterns of cutaneous T-cell lymphomas in the United States. *Leuk Lymphoma*. 2013; 54(4): 752–759.
6. Argnani L, Broccoli A, Zinzani PL. Cutaneous T-cell lymphomas: Focusing on novel agents in relapsed and refractory disease. *Cancer Treat Rev*. 2017; 61, 61–69.

Presented at ISPOR, 2022

Sponsored by Kyowa Kirin, Inc.