Real-world Healthcare Resource Utilization and Associated Costs in Patients With Eosinophilic Esophagitis

EE497

Tom Tencer,[1] Qian Xia,[1] Greeta Jobson,[2] Ellen Qian,[1] Evan S. Dellon[3]

[1]Bristol Myers Squibb, Princeton, NJ, USA; [2]Mu Sigma, Bengalaru, India; [3]Center for Esophageal Diseases and Swallowing Division of Gastroenterology and Hepatology, Department of Medicine, University of North Carolina School of Medicine, Chapel Hill, NC, USA

Introduction

- Eosinophilic esophagitis (EoE) is a chronic, immune/Th2 inflammatory esophageal disease characterized clinically by symptoms related to esophageal dysfunction and histologically by eosinophil-predominant inflammation[1]
 - Pathogenesis of EoE is associated with familial inheritance and environmental exposures[2]
 - From a worldwide systematic review, including studies from 2004 to 2019, the estimate of overall prevalence for patients with EoE is 34.2/100,000 inhabitants[3]
 - In the US, estimated overall prevalence of EoE ranges from 25.9 to 56.7/100,000 (2001-2015) and for those individuals, most are male and of Caucasian origin[4-7]
 - Worldwide incidence and prevalence increased from 2004 to 2019[3-8]
- Chronic inflammation contributes to tissue remodeling, fibrosis, and stricture formation[9]; this inflammation results in a range of symptoms including dysphagia, food impaction, abdominal pain, and symptoms of gastroesophageal reflux disease,[1] resulting in a major cause of gastrointestinal morbidity and associated financial burden[10]
- EoE is associated with high economic burden in the US
 - Total annual median healthcare costs in 2010 per case vs control: $3304 vs $1001 ($2302 EoE-attributable)[11]
 - Estimated total annual costs in 2010: $503 million to $1.4 billion ($350–$947 million EoE-related excess costs)[11]
- Few real-world studies have investigated healthcare resource utilization (HCRU) and associated costs[12]

Objective

- To compare HCRU and costs in patients with EoE and matched controls without EoE

Methods

Study design

- This matched case-control study used US medical and pharmacy claims from OPTUM Clinformatics Data Mart (1/1/2008–9/30/2020) to identify patients with EoE (cases) and individuals without EoE (controls) and their associated HCRU and costs (**Figure 1**)
- Index date was defined as the date of first EoE diagnosis (index period: 1/1/2009–9/30/2019) and follow-up was 12-months post index
- Individuals in the control cohort were assigned index dates so that the length of time from the eligibility start date to the index date for both cases and control was same

Figure 1. Study design

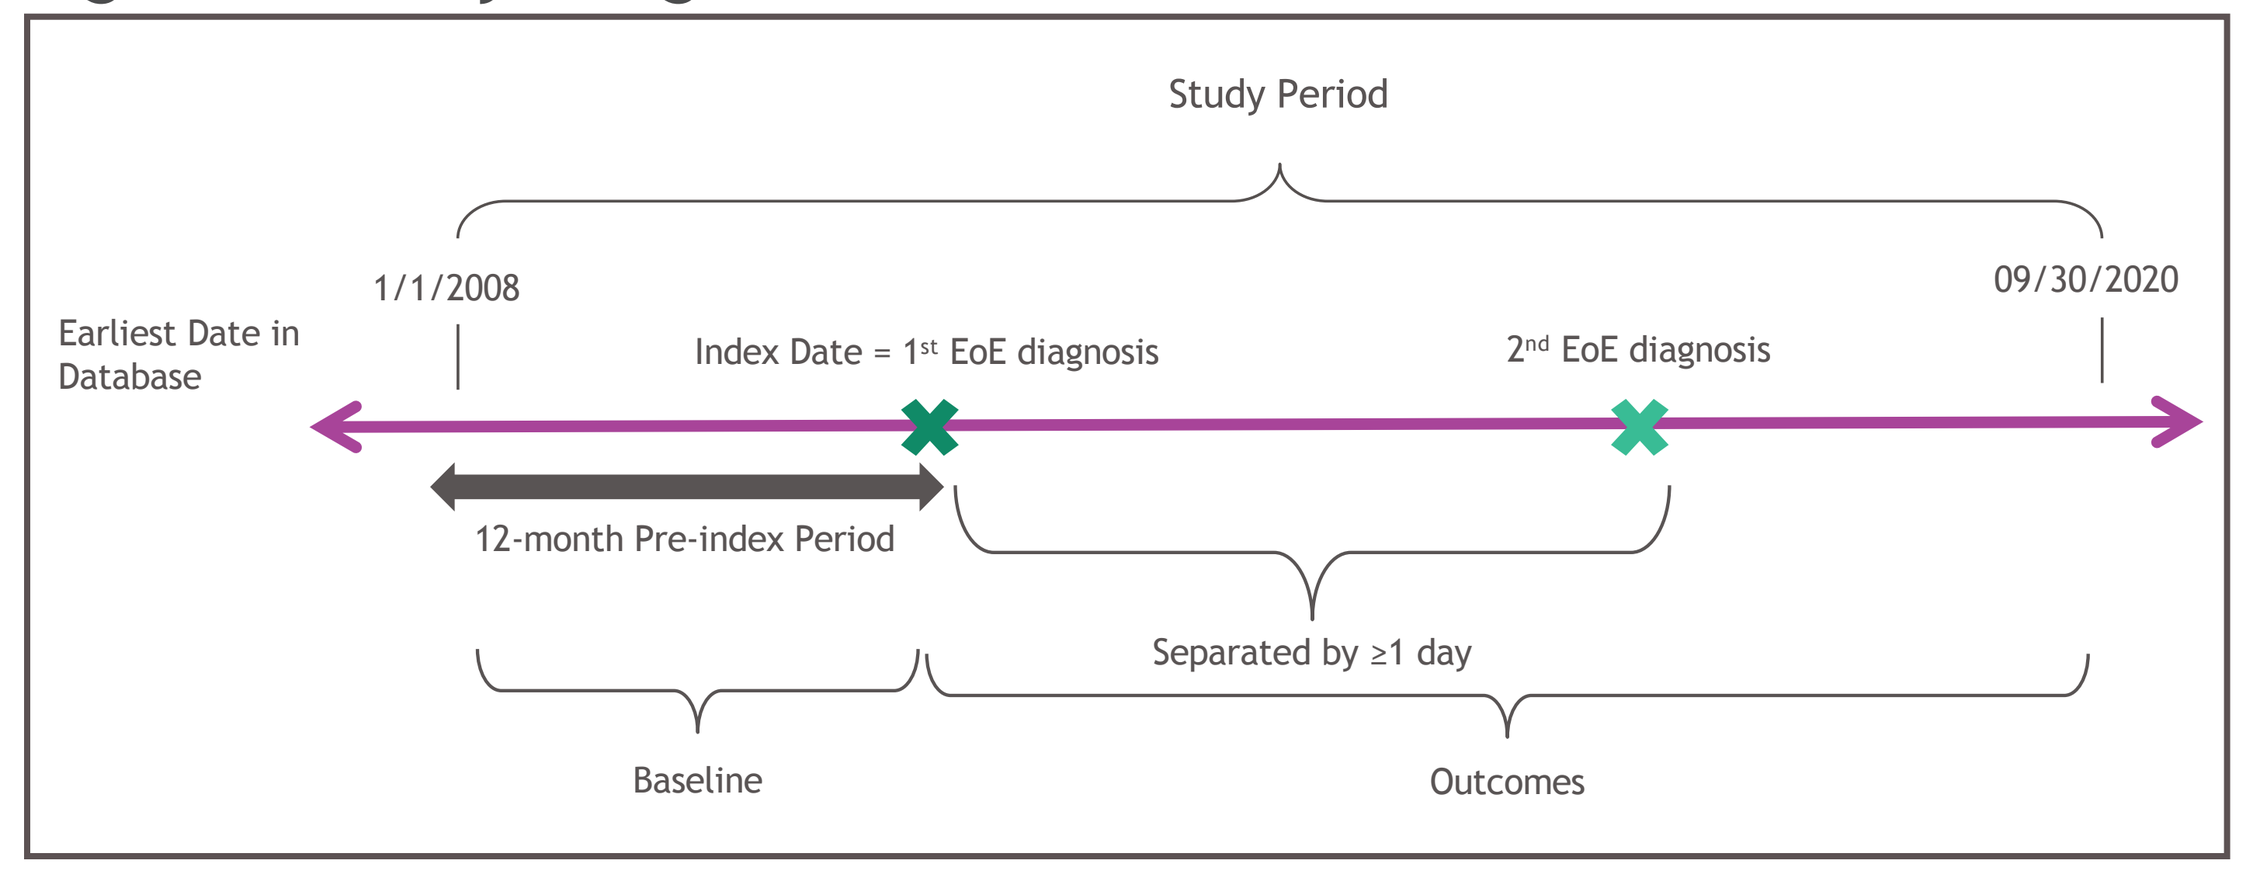

Study Period
Earliest Date in Database
1/1/2008
09/30/2020
Index Date = 1st EoE diagnosis
2nd EoE diagnosis
12-month Pre-index Period
Separated by ≥1 day
Baseline
Outcomes

Study period: January 1, 2008 to September 30, 2020.
Index period: January 1, 2009 to September 30, 2019.
Baseline period: 12 months before index date – 1st EoE diagnosis.
Follow-up: 12 months post-index date.
EoE, eosinophilic esophagitis.

Cohort selection

Inclusion criteria

- Patients with at least 1 claim with an EoE diagnosis (ICD-9-CM code: 530.13; ICD-10-CM code: K20.0) during the index period
- Patients with ≥2 claims with an EoE diagnosis on 2 different dates (separated by at least 1 day)
- Patients continuously enrolled in a health plan (both medical and prescription drug coverage) for at least 12 months before and after the index date

Exclusion criteria

- Patients with at least 1 claim with an oropharyngeal or esophageal cancer diagnosis at any time during the study period
- Patients with a diagnosis of any malignancy at any time during the study period

Matched cohorts

- Cases (with EoE)
- Controls (without EoE): a random sample of enrollees in the database were identified from a group of people at risk of EoE (ie, the cases were also allowed to be controls until they had a diagnosis of EoE)
- Cases were matched to controls in a 1:4 ratio by: sex, age, geographic region (as of the index date), Charlson comorbidity index (CCI) classification during the pre-index period, and the cohort entry date (±120 days), so that they enter the cohort around the same time and length of continuous enrollment from the eligibility start date to the index date to make sure controls are still at risk for having the outcome event after the same number of days after cohort entry as the case (**Figure 2**)

Figure 2. Cohort selection

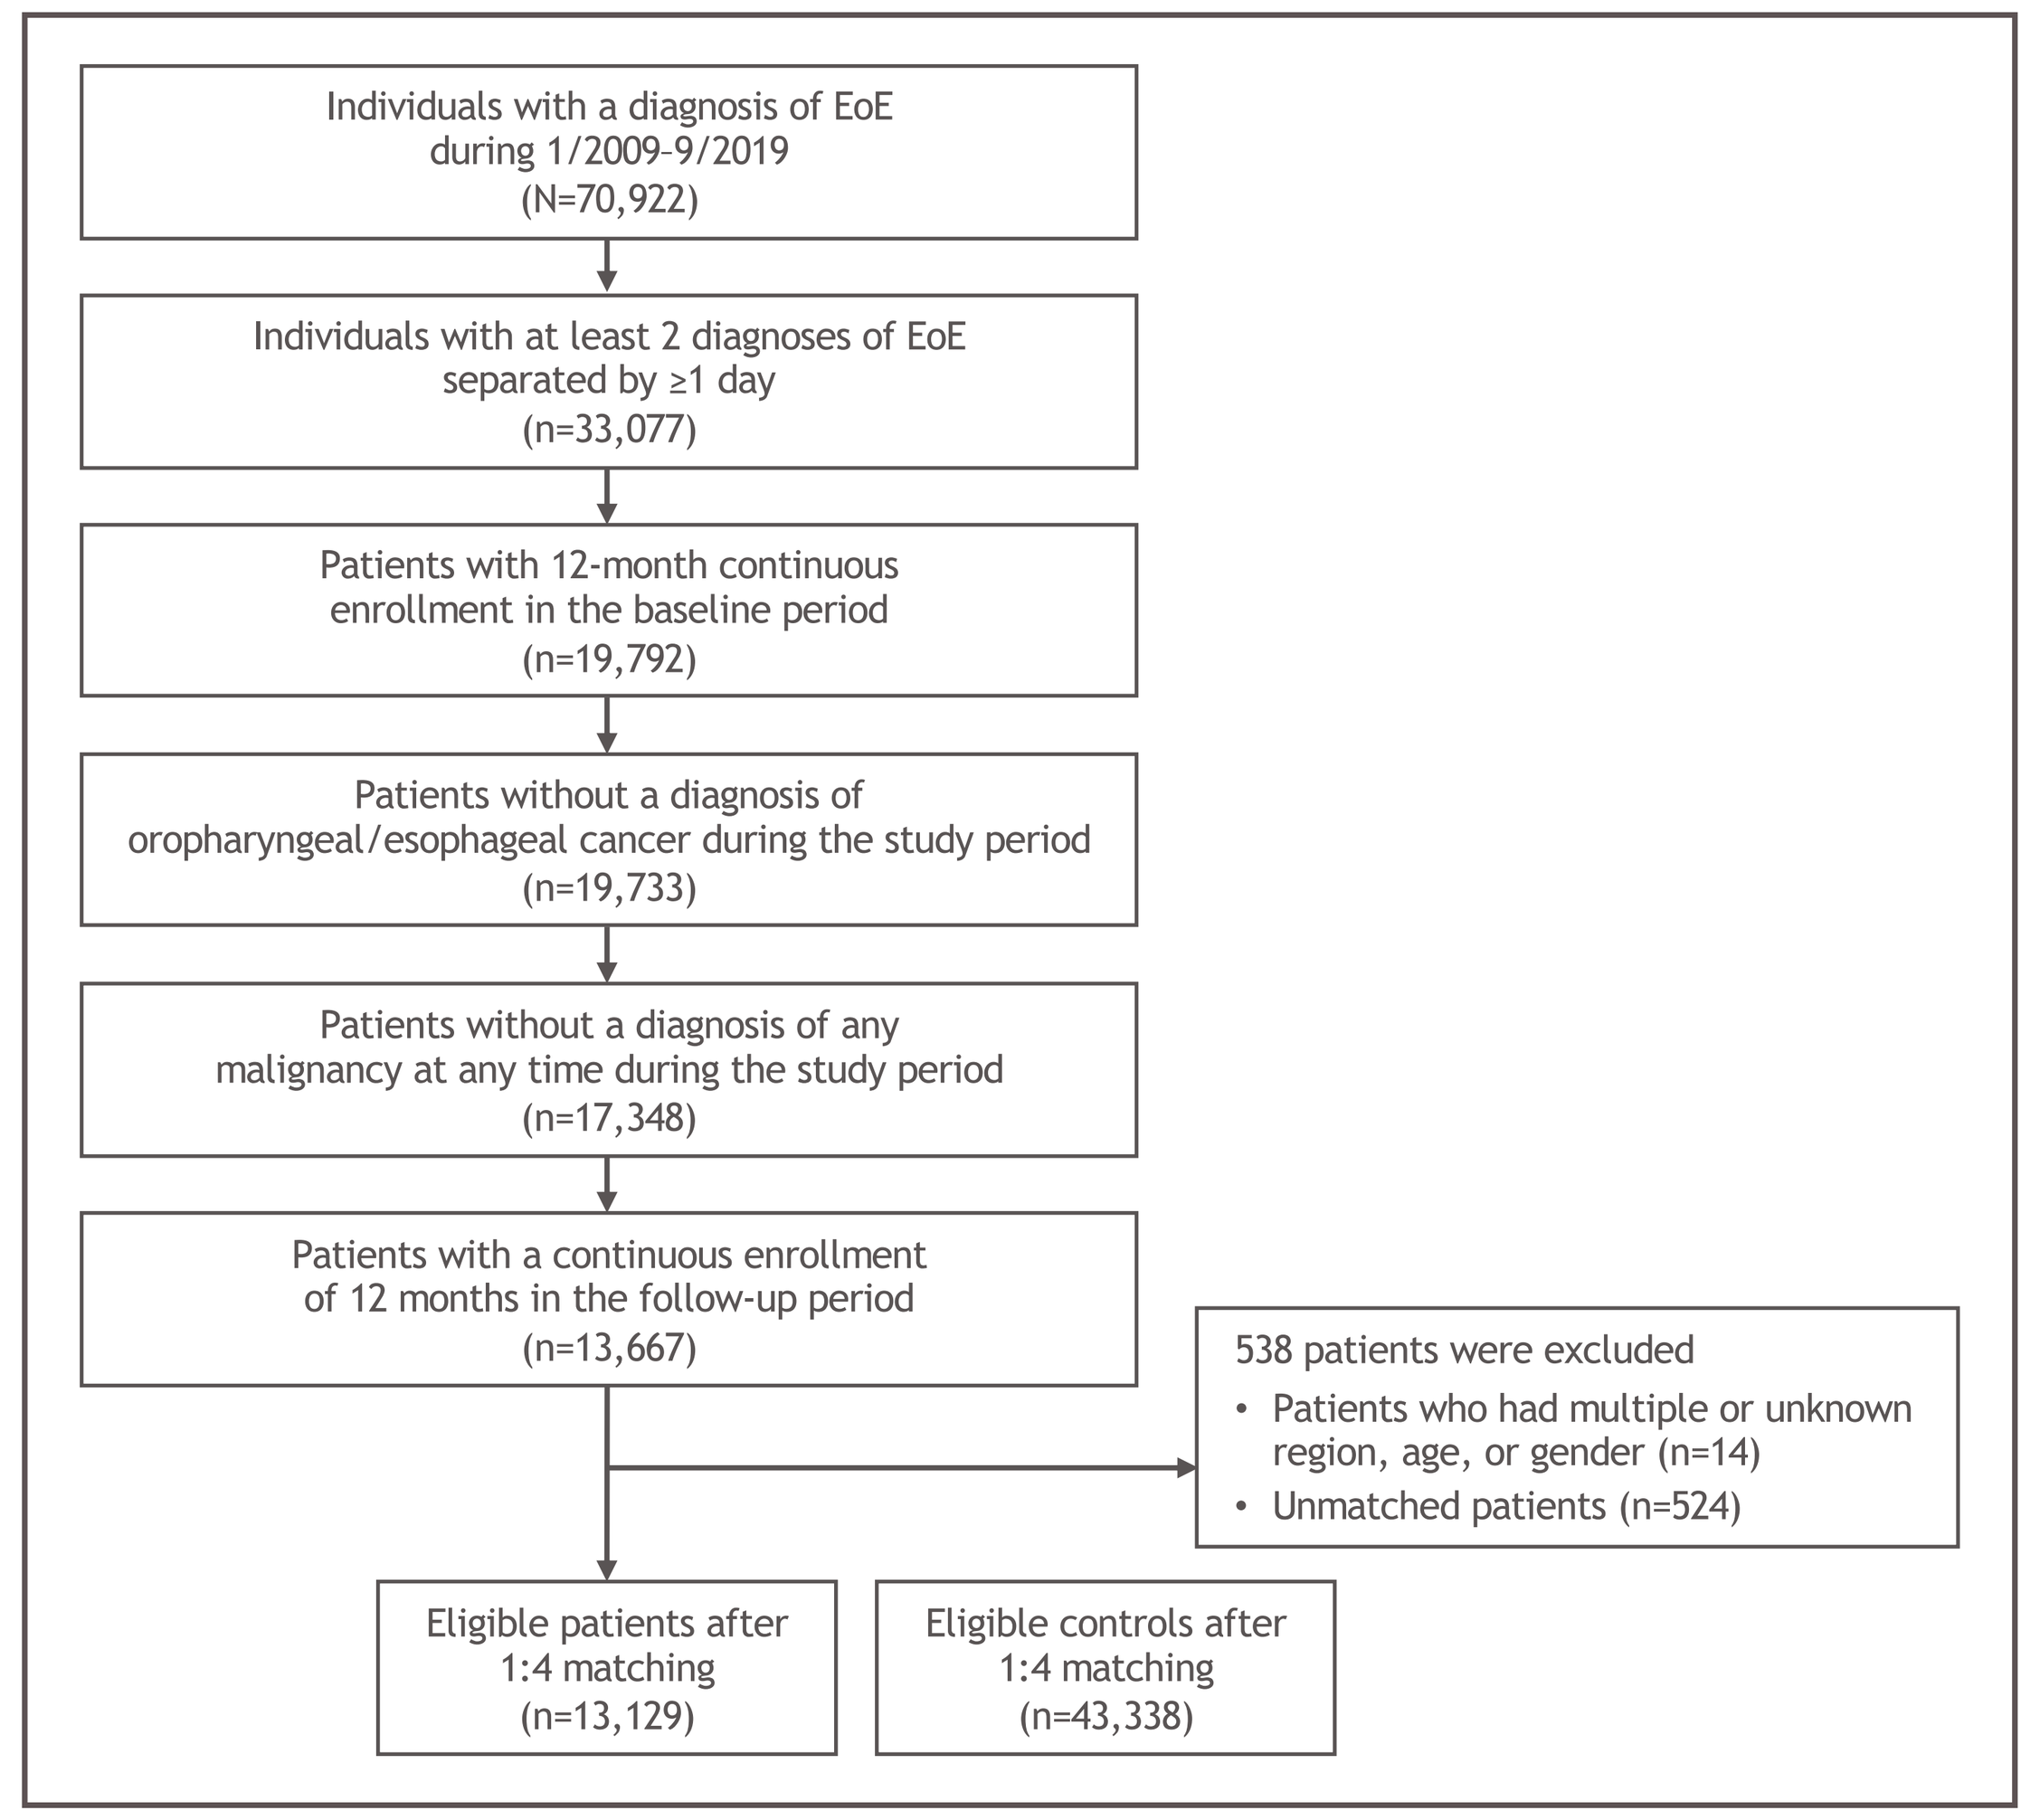

Individuals with a diagnosis of EoE during 1/2009-9/2019 (N=70,922)
Individuals with at least 2 diagnoses of EoE separated by ≥1 day (n=33,077)
Patients with 12-month continuous enrollment in the baseline period (n=19,792)
Patients without a diagnosis of oropharyngeal/esophageal cancer during the study period (n=19,733)
Patients without a diagnosis of any malignancy at any time during the study period (n=17,348)
Patients with a continuous enrollment of 12 months in the follow-up period (n=13,667)
538 patients were excluded
• Patients who had multiple or unknown region, age, or gender (n=14)
• Unmatched patients (n=524)
Eligible patients after 1:4 matching (n=13,129)
Eligible controls after 1:4 matching (n=43,338)

EoE, eosinophilic esophagitis.

Outcomes

- All-cause HCRU and associated costs (settings: inpatient [IP] and outpatient [OP] medical services, emergency room [ER] and pharmacy) during baseline and follow-up
- All-cause costs by treatment pattern (persistence, re-initiation, pure discontinuation, and switch) and setting (**Table 1**)
- All-cause costs pre- and post-treatment switch by setting

Table 1. Treatment pattern terminology

Terminology	Definition
Persistence	Continuous treatment episode without allowable treatment gaps from the previous treatment
Switch	Initiation of a new treatment before the last day of supply of the previous treatment or within the allowable gap OR If a patient moves to a different drug class after discontinuing from the previous treatment after the allowable gap during follow-up
Pure discontinuation	No other treatment following >90-day gap from the previous treatment
Re-initiation	Treatment gap >90 days from the previous treatment, followed by the same treatment
Allowable treatment gap	Days' supply + 90 days

Statistical analysis

Descriptive statistics for patient characteristics and outcomes

- Continuous variables summarized using means and standard deviations
- Categorical variables presented as counts and percentages

All-cause HCRU and costs (IP, OP, ER, pharmacy) at baseline and follow-up: 1000 patients per month (1000PPM) (HCRU) and per-patient-per-month (PPPM) (cost)

- Costs were inflation-adjusted to 2020 using the medical care component of the consumer price index

HCRU and costs compared: EoE (cases) vs without EoE (matched controls)

- Analyses were also stratified by treatment pattern
- Generalized linear model was used to assess the relationship between cohorts (cases vs matched controls) and total HCRU and costs, adjusted for baseline patient characteristics
- *P* value ≤0.05 was considered statistically significant

Results

Baseline characteristics

- A total of 13,129 EoE cases and 43,338 matched controls were included in this study (**Table 2**)
- Mean age across cohorts was approximately 37 years and most patients (~65%) were male

Table 2. Baseline characteristics

Patient Characteristics at Baseline	EoE cases	Controls
Total patients, N	13,129	43,338
Age at index date, mean (SD)	37.55 (18.61)	37.33 (19.01)
Male, n (%)	8462 (64.45)	28,109 (64.86)
Region, n (%)		
Midwest	4123 (31.40)	13,878 (32.02)
Northeast	1150 (8.76)	3676 (8.48)
South	4799 (36.55)	15,592 (35.98)
West	3042 (23.17)	10,135 (23.39)
Other	15 (0.11)	57 (0.13)
EoE-related comorbidity, n (%)		
GERD[a]	5096 (38.81)	2458 (5.67)
Allergic rhinitis	2704 (20.60)	3533 (8.15)
Asthma	2079 (15.84)	2214 (5.11)
Eczema	387 (2.95)	466 (1.08)
Food allergy	749 (5.70)	184 (0.42)
Complications of EoE		
Esophageal stricture	1565 (11.92)	60 (0.14)
Food impaction	606 (4.62)	7 (0.02)
CCI mean (SD)	0.28 (0.59)	0.15 (0.45)
Prior treatments, n (%)		
Esophageal dilation	771 (5.87)	49 (0.11)
Patients taking medications of interest	7180 (54.69)	9049 (20.88)

[a]GERD is often diagnosed in patients prior to EoE diagnosis and is a part of differential diagnosis; therefore, GERD may not be a comorbidity.
CCI, Charlson Comorbidity Index; EoE, eosinophilic esophagitis; GERD, gastroesophageal reflux disease; SD, standard deviation.

All-cause HCRU (per 1000PPM) and costs ($PPPM)

- EoE cases had more HCRU visits across all settings versus controls (1986.2 vs 1168.4 per 1000PPM) and higher costs ($800.39 vs $402.81 PPPM) at baseline and at 12 months (visits: 2535.8 vs 1220.3 per 1000PPM; costs: $1184.73 vs $463.25 PPPM)
- Across settings, most HCRU and costs were higher during follow-up than at baseline
- Outpatient vists were a significant driver of resource use and costs during baseline and follow-up (**Figure 3**)

Figure 3. All-cause HCRU (Per 1000PPM) and costs ($PPPM)

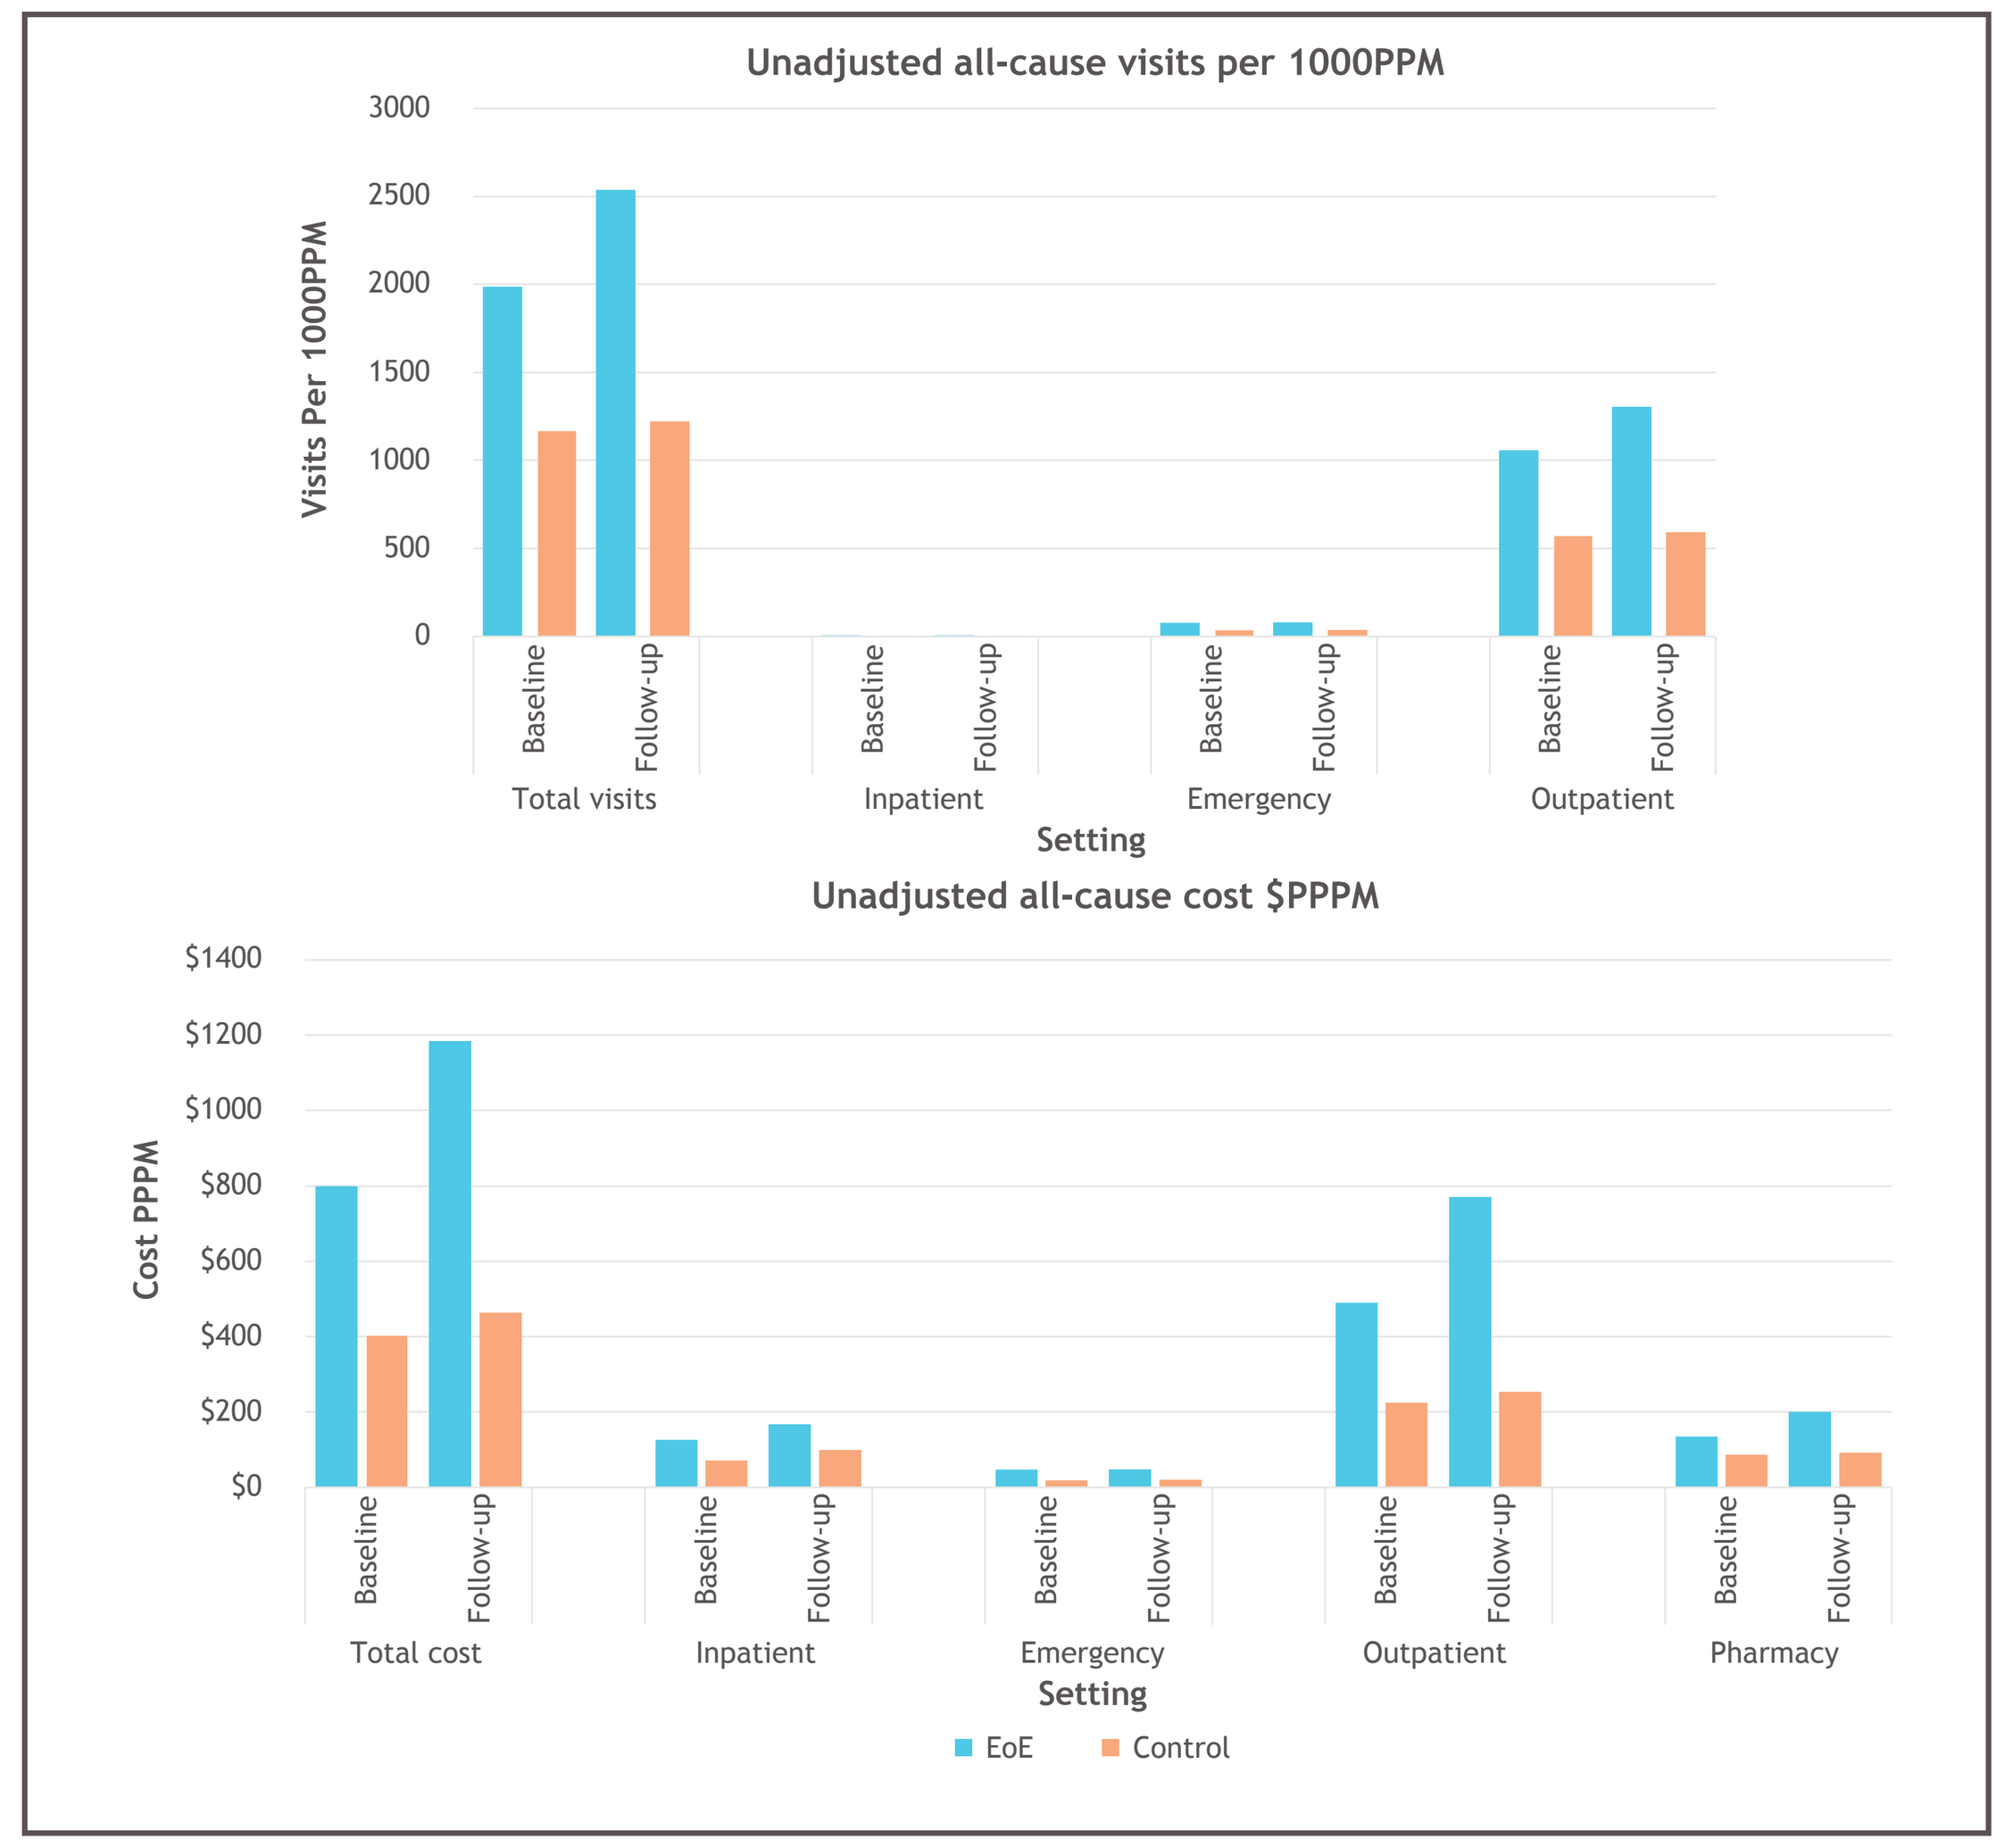

Unadjusted all-cause visits per 1000PPM
Visits Per 1000PPM
3000
2500
2000
1500
1000
500
0
Baseline
Follow-up
Total visits
Inpatient
Emergency
Outpatient
Setting
Unadjusted all-cause cost $PPPM
Cost PPPM
$1400
$1200
$1000
$800
$600
$400
$200
$0
Total cost
Inpatient
Emergency
Outpatient
Pharmacy
Setting
EoE
Control

1000PPM, 1000 patients per month; EoE, eosinophilic esophagitis; PPPM, per-patient-per-month.

Adjusted all-cause HCRU and costs

- EoE was associated with 388.8 more outpatient visits, 14.7 more ER visits, and 2.8 more inpatient visits (per 1000PPM) (**Table 3**)
- Incremental medical costs for EoE cases were $581.15 PPPM, primarily driven by outpatient costs ($349.22 PPPM)
- Pharmacy ($75.34 PPPM), inpatient ($57.51 PPPM), and ER ($10.67 PPPM) costs also were higher for EoE cases
- All incremental costs were statistically significant

Table 3. Adjusted all-cause HCRU and costs

Settings	EoE (95% CI)	Control (95% CI)	Incremental cost/HCRU (95% CI)
HCRU, visits per 1000PPM			
Inpatient	6.74 (0.20, 9.51)	3.99 (0.12, 5.81)	2.75 (0.09,3.97)*
Emergency room	34.62 (10.38, 73.39)	19.90 (5.99, 42.39)	14.73 (4.28, 32.07)*
Outpatient	891.89 (834.03, 954.99)	503.08 (468.34, 538.93)	388.81 (362.10, 417.98)*
Costs, $PPPM			
Inpatient	$108.79 ($42.11, $196.82)	$51.28 ($18.67, $95.38)	$57.51 ($22.14, $103.05)*
Emergency room	$20.89 ($15.16, $26.75)	$10.22 ($7.39, $13.39)	$10.67 ($7.32, $13.83)*
Outpatient	$508.18 ($448.69, $565.20)	$158.95 ($140.62, $177.50)	$349.22 ($309.08, $391.24)*
Pharmacy	$114.32 ($92.09, $132.42)	$38.98 ($31.25, $46.10)	$75.34 ($60.32, $87.35)*
Total	**$908.45 ($656.64, $936.00)**	**$327.30 ($233.33, $339.41)**	**$581.15 ($420.83, $600.09)***

*P<0.001.
1000PPM, 1000 patients per month; CI, confidence interval; EoE, eosinophilic esophagitis; HCRU, healthcare resource utilization; $PPPM, cost per patient per month.

All-cause costs by treatment pattern

- Outpatient costs accounted for the greatest proportion of costs among settings and treatment patterns (**Figure 4A**)
- Switchers ($1388.49 PPPM) had the highest all-cause costs and pure discontinuers ($869.55 PPPM) had the lowest among treatment patterns
- Among switchers, all-cause costs were lower after switch in all settings (**Figure 4B**)

Figure 4. All-cause costs PPPM among treatment patterns across settings

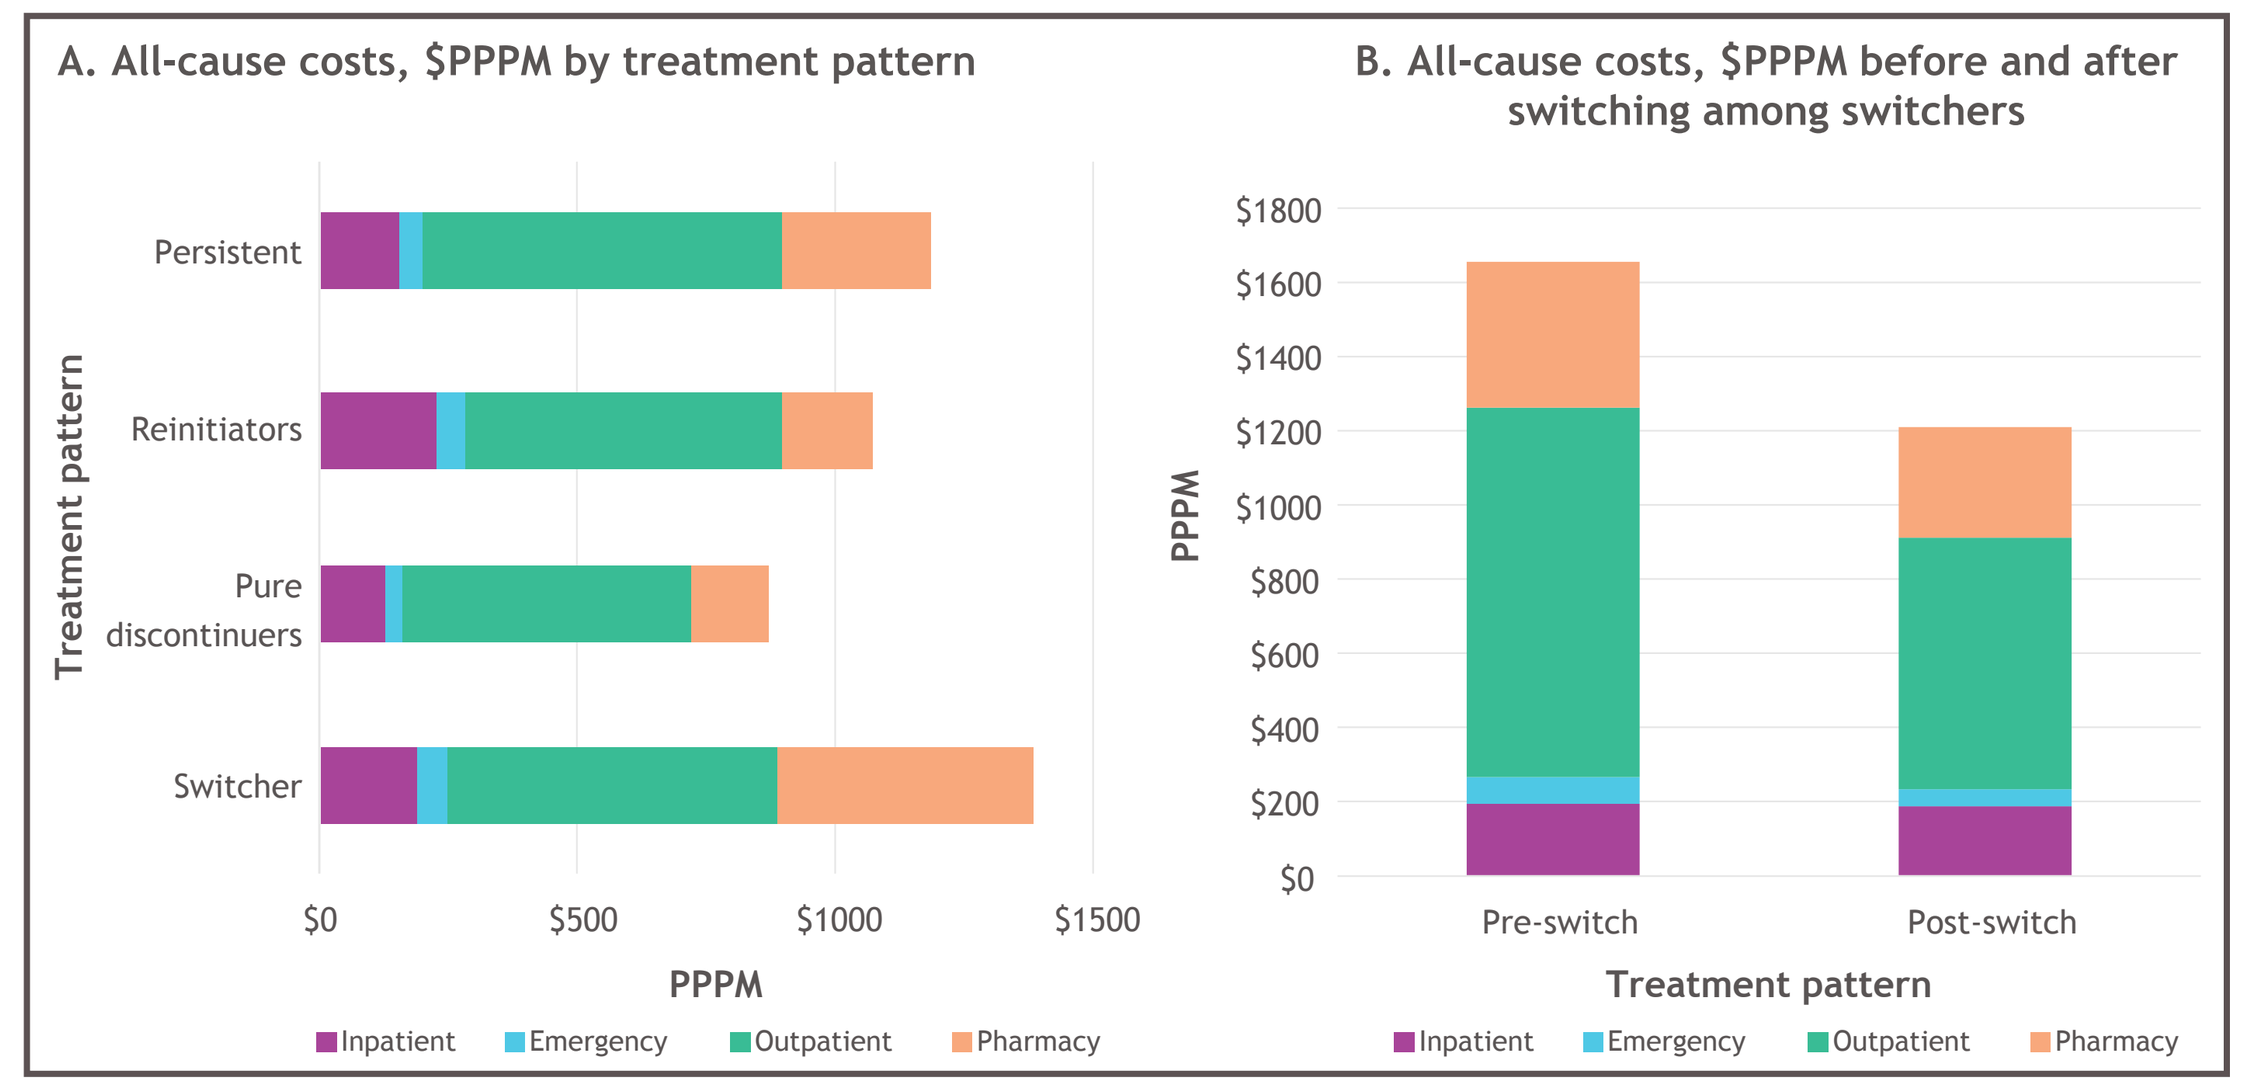

A. All-cause costs, $PPPM by treatment pattern
Persistent
Reinitiators
Pure discontinuers
Switcher
Treatment pattern
$0
$500
$1000
$1500
PPPM
Inpatient
Emergency
Outpatient
Pharmacy
B. All-cause costs, $PPPM before and after switching among switchers
$1800
$1600
$1400
$1200
$1000
$800
$600
$400
$200
$0
PPPM
Pre-switch
Post-switch
Treatment pattern
Inpatient
Emergency
Outpatient
Pharmacy

PPPM, per-patient-per-month.

Limitations

- Claims data are collected primarily for billing and reimbursement purposes and therefore may incompletely capture medical conditions that are found in medical records; medical coding is imperfect and may lead to misclassification bias due to misdiagnosis or coding errors
- Claims databases only capture data from patients covered by the health plan; therefore, results may not be generalizable to all individuals with EoE

Conclusions

- EoE was associated with greater economic burden compared with matched controls, driven primarily by outpatients visits
- Treatment switching in patients with EoE was associated with greater costs compared with other treatment patterns, suggesting a need for more optimal treatments in EoE

References

1. Dellon ES, et al. *Gastroenterology*.2018;155:1022-33. **2.** Furuta GT, Katzka DA. *N Engl J Med*. 2015;373:1640-8. **3.** Navarro P, et al. *Aliment Pharmacol Ther*. 2019;49:1116-25. **4.** Maradey-Romero C, et al. *Aliment Pharmacol Ther*. 2015;41:1016-22. **5.** Dellon ES, et al. *Clin Gastro and Hep*. 2014;12:589-96. **6.** Mansoor E, Cooper GS. *Dig Dis Sci*. 2016;61:2928-34. **7.** Arias Á, Lucendo AJ. *Exp Rev Gastroenterol Hepatol*. 2020;14:1069-82. **8.** Lucendo AJ, et al. *Ther Adv Gastroenterol*. 2022;15:17562848221074204. **9.** Armbruster-Lee J, et al. *J Leukoc Biol*. 2017;104:31-40. **10.** Dellon ES, Hirano I. *Gastroenterology*. 2018;154:319-32. **11.** Jensen ET, et al. *Am J Gastroenterol*. 2015;110: 626-32. **12.** Dellon ES. *Ann Allergy Asthma Immunol*. 2019;123:166-72.

Acknowledgments

- This study was sponsored by Bristol Myers Squibb.
- Professional medical writing from LeeAnn Braun, MPH, MEd and editorial assistance were provided by Peloton Advantage, LLC, an OPEN Health company, Parsippany, NJ, USA, and were funded by Bristol Myers Squibb.

Relationships and Activities

- **TT:** Employee of and stock ownership in Bristol Myers Squibb.
- **QX:** Employee of and stock ownership in Bristol Myers Squibb.
- **GJ:** Employee of Mu Sigma, which has received consulting fees from Bristol Myers Squibb.
- **EQ:** Fellow with Bristol Myers Squibb.
- **ESD:** Received grant or contract support from Adare/Ellodi, Allakos, Arena, AstraZeneca, GSK, Meritage, Miraca, Nutricia, Celgene/Receptos/BMS, Regeneron, Revolo, and Shire/Takeda; educational grants from Allakos, Banner, and Holoclara; and consulting fees from Abbot, AbbVie, Adare/Ellodi, Aimmune, Akesobio, Allakos, Amgen, Arena, AstraZeneca, Avir, Biorasi, Calypso, Celgene/Receptos/BMS, Celldex, Eli Lilly, EsoCap, GSK, Gossamer Bio, Invea, Landos, LucidDx, Morphic, Nutricia, Parexel/Calyx, Phathom, Regeneron, Revolo, Robarts/Alimentiv, Salix, Sanofi, Shire/Takeda, and Target RWE.

Presented at ISPOR 2022, May 15–18, 2022, Washington, DC, Area and Virtual

Email: Qian.Xia@bms.com

Copies of this poster are for personal use and may not be reproduced without written permission from the authors.