CLARIO.

At-home versus in-clinic patient reported outcome compliance in oncology clinical trials

Dumais K, Ph.D.[1], McDowell B[2]
[1] Clario, Medford, MA, USA, [2] Clario, Geneva, Switzerland

Introduction

- There is increased regulatory emphasis on using patient reported outcome (PRO) assessments in oncology clinical trials to provide additional information on clinical benefit beyond traditional survival and tumor response endpoints.[1-4]
- The latest oncology recommendations from the FDA outlines core PROs to consider and highlights the need for more frequent assessments to obtain a more accurate depiction of patients' disease and treatment-related symptoms and quality of life.[4]
- To accommodate recommendations for more frequent assessments, which may be weekly or biweekly assessments for the first few treatment cycles, there has been a trend for PROs to be completed by patients at home versus in the clinic. This allows for more frequent collection while reducing the burden of patient travel and site visit time.
- However, given disease progression characteristics, oncology studies are already at risk for low compliance, and unsupervised at-home PRO collection puts greater responsibility on patients.
- **Objective:** We aimed to determine the impact of at-home PRO collection versus at-site PRO collection on PRO completion compliance.

Methods

- We analyzed data from 25 oncology clinical trials for which Clario provided electronic clinical outcome assessment (eCOA) services. Studies varied across clinical phase, cancerous condition, number of PROs, PROs used and PRO administration schedule. Analysis included studies that collected PROs via a handheld device (with most or all assessments completed at patients' homes; N=17 studies) or via a tablet device (with all assessments completed at a clinical trial site; N=8 studies). Analysis included 849,091 administered PRO assessments across 14,284 participants.
- We compared the number of PROs used and the PRO completion compliance rate (i.e., received PRO forms versus missed PRO forms) in studies collecting PROs via a handheld device (home PRO completion) versus studies collecting PROs via a tablet device (site PRO completion), via two-tailed two-sample t-test with equal variance. We also correlated the number of PROs per study with PRO completion compliance rates using Pearson's correlation.

Results

The number of PROs per study did not differ between studies using a handheld device at home (M= 5.8, SD= 1.5, range = 3-8) versus a tablet device at site (M= 4.8, SD= 1.7, range 2-7; t(23)= 1.6, P= .12; Figure 1).

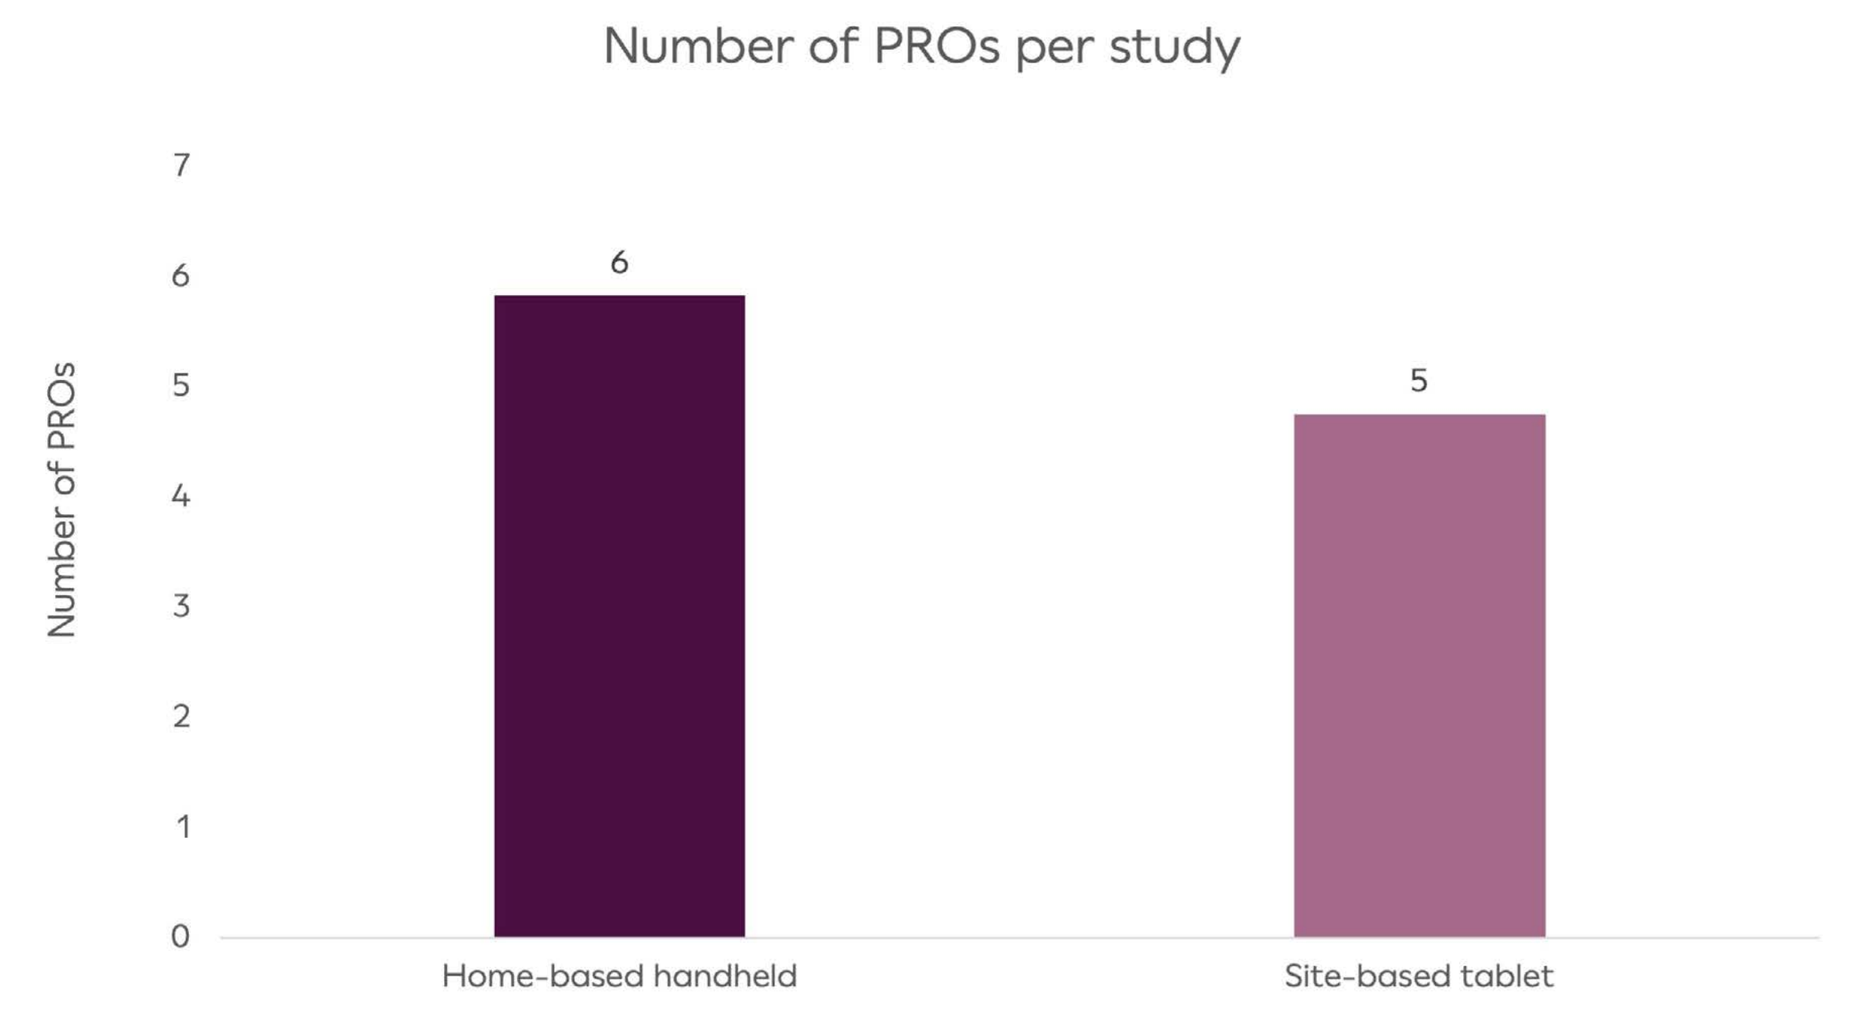
Number of PROs per study
Number of PROs
0
1
2
3
4
5
6
7
6
5
Home-based handheld
Site-based tablet

Figure 1. Average number of PROs per study for studies using a handheld device at home versus a tablet device at site.

PRO completion compliance did not differ between studies collecting PROs via a handheld device at home (M= 83.9%, SD= 6.6%, range 72-93%) versus studies using a tablet device at site (M= 86.3%, SD=7.3%, range 76-94%; t(23)= -0.8, P= .44; Figure 2).

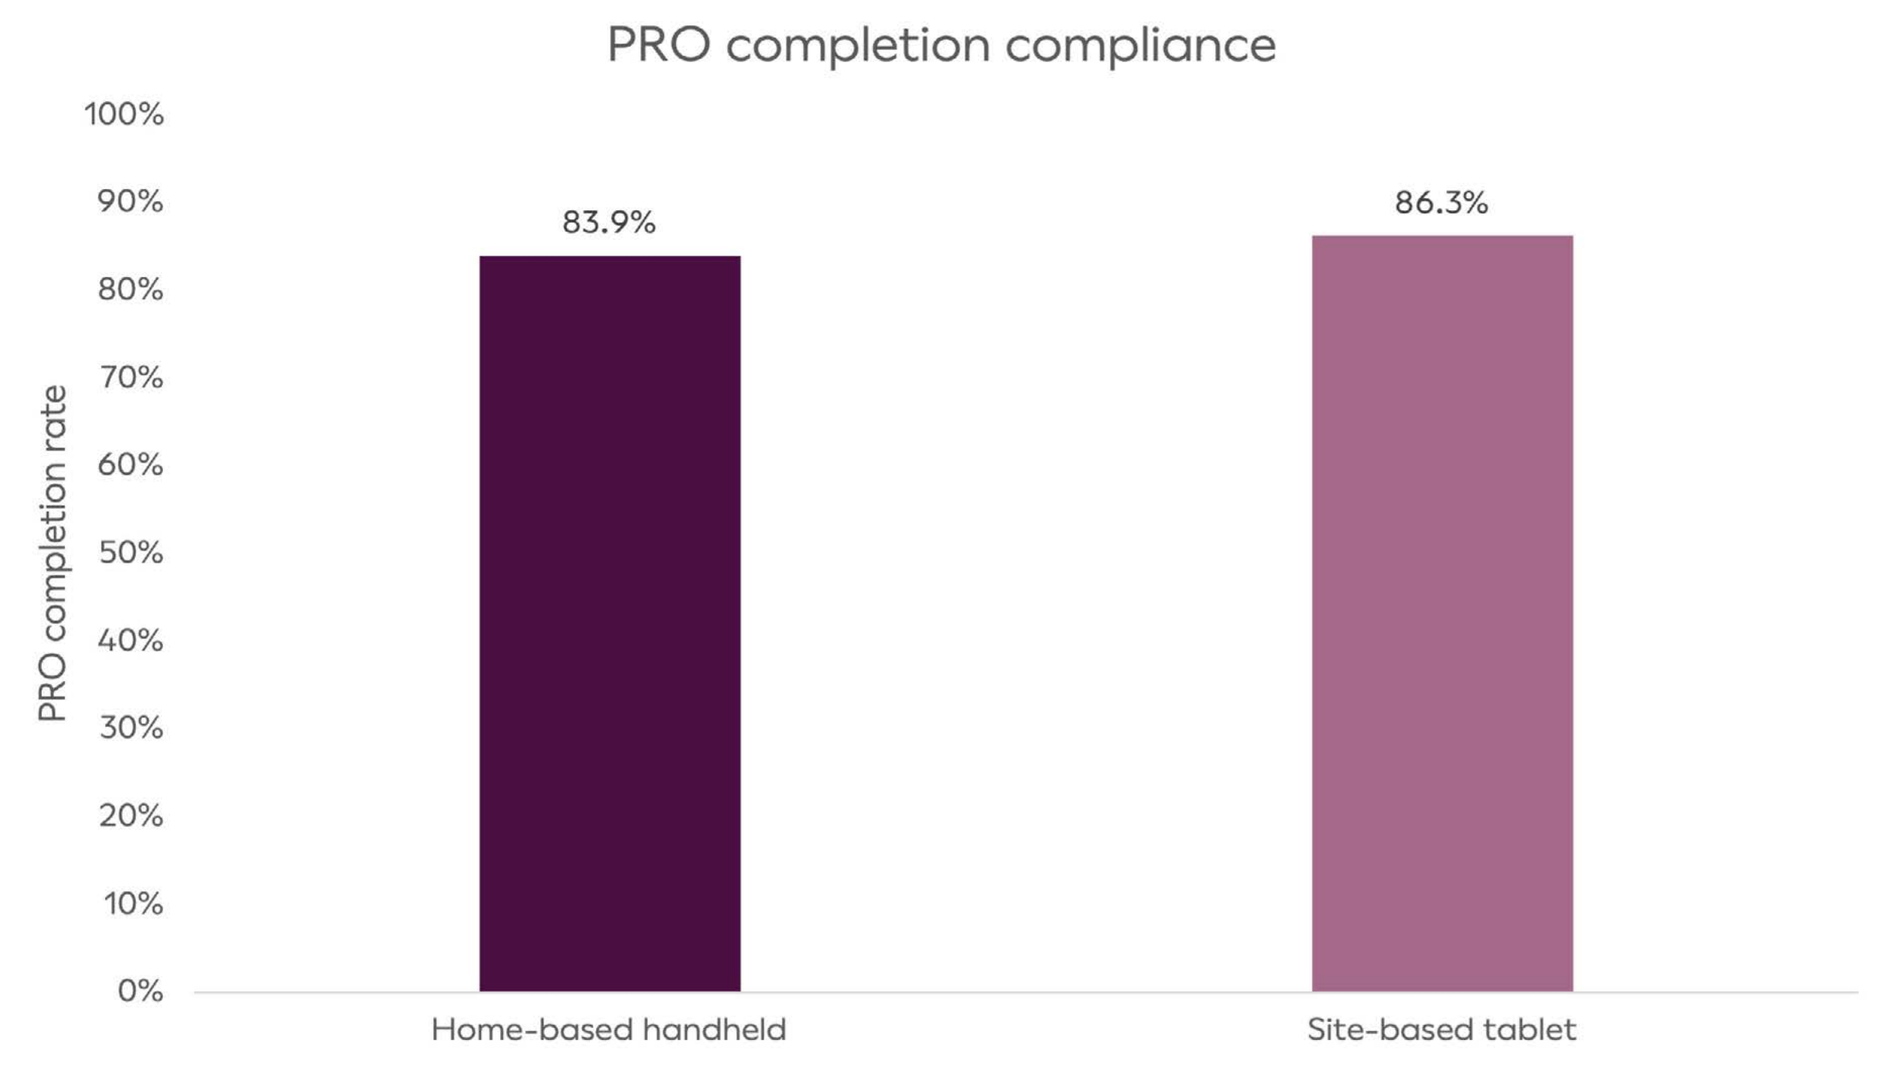
PRO completion compliance
PRO completion rate
0%
10%
20%
30%
40%
50%
60%
70%
80%
90%
100%
83.9%
86.3%
Home-based handheld
Site-based tablet

Figure 2. Average PRO completion rate for studies using a handheld device at home versus a tablet device at site.

The PRO completion compliance rate did not correlate with the number of PROs per study (r(23)= -.004, P= .98; Figure 3).

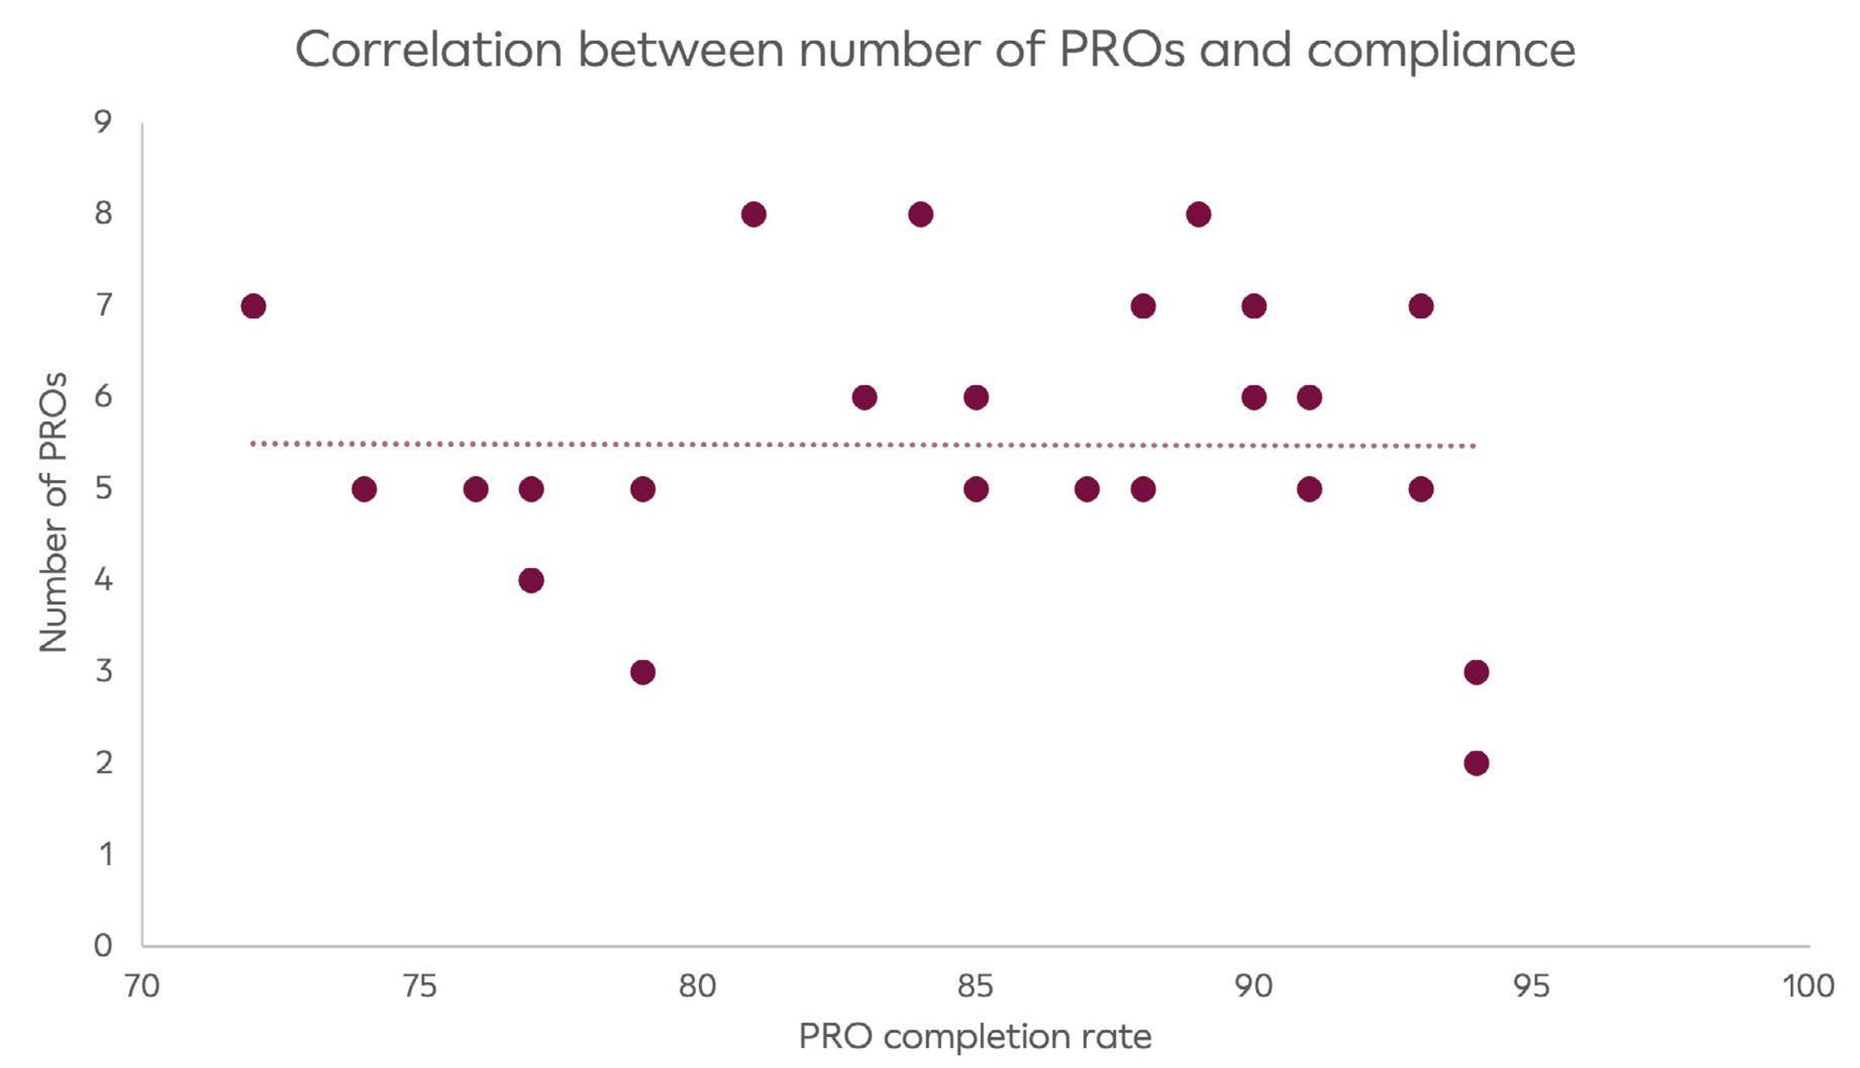
Correlation between number of PROs and compliance
Number of PROs
0
1
2
3
4
5
6
7
8
9
70
75
80
85
90
95
100
PRO completion rate

Figure 3. Correlation between number of PROs per study and PRO completion compliance rate for all studies.

Conclusion

- With new regulatory guidance on PROs in oncology comes the responsibility of those designing and conducting trials to not only collect what is meaningful to patients, but to do so in a way that minimizes patient burden. In considering new recommendations for more frequent PRO administration, at-home data collection strategies may reduce patient burden by reducing patient travel and visit time.
- Our results show that PRO compliance remains high when collected at home on a handheld device, suggesting that at-home electronic PRO completion is a viable and useful strategy for PRO collection in oncology clinical trials, and does not negatively impact PRO compliance rate.
- Given recent regulatory emphasis on PROs in oncology trials, there is potential for more PROs to be collected per study in order to gain a more thorough and accurate assessment of patients' health-related quality of life. In considering the number of PROs used in a study, the impact on patient burden and patients' willingness or motivation to complete a large number of PROs is an important factor.
- Our results show that the number of PROs used in a study does not correlate with PRO compliance rate, indicating that even those studies that include a larger number of PROs show high compliance. This builds on previous studies showing that oncology patients want to report their symptoms, indicate significant benefit in doing so and report minimal response burden upon completing a large battery of PRO measures.[5,6]

References

1. EMA Guideline on the clinical evaluation of anticancer medicinal products, Jan 2019. https://www.ema.europa.eu/en/evaluation-anticancer-medicinal-products-man#current-version---under-revision--section. Retrieved April 1, 2022.
2. EMA Appendix 2 to the guideline on the evaluation of anticancer medicinal products in man - the use of patient-reported outcome (PRO) measures in oncology studies, April 2016. https://www.ema.europa.eu/en/appendix-2-guideline-evaluation-anticancer-medicinal-products-man-use-patient-reported-outcome-pro. Retrieved April 1, 2022.
3. FDA Guidance for Industry: Clinical trial endpoints for the approval of cancer drugs, Dec 2018. https://www.fda.gov/regulatory-information/search-fda-guidance-documents/clinical-trial-endpoints-approval-cancer-drugs-and-biologics. Retrieved April 1, 2022.
4. FDA Draft Guidance for Industry: Core Patient-Reported Outcomes in Cancer Clinical Trials, June 2021. https://www.fda.gov/regulatory-information/search-fda-guidance-documents/core-patient-reported-outcomes-cancer-clinical-trials. Retrieved April 1, 2022.
5. Peechatka AL, Gerzon, M, Ly, JJ, Dallabrida SM, 2020. Oncology patient preferences in reporting on symptoms. Int J of Clinical Trials 7(1):11-17.
6. Atkinson TM, Schwartz CE, Goldstein L, Garcia I, Storfer DF, Li Y, Zhang J, Bochner BH, Rapkin BD, 2019. Perceptions of response burden associated with completion of patient-reported outcome assessments in oncology. Value Health 22(2):225-230.

ISPOR | May 15–18, 2022