Systemic antineoplastic treatment utilization among patients with advanced HER2+ breast cancer across large community health systems in the US

Lorenzo R, Pomerantz D, Wolf F, Sommers C, Brown T, Rioth M, Wolfe T, Lerman M, Sanchez N
Syapse, San Francisco CA, United States

Syapse®
ISPOR 2022

OBJECTIVES

Community health systems (CHS) in the US play a large role in the care of patients with cancer, with over 50% of patients with cancer cared for in CHS.

The systemic treatments used among patients with advanced HER2+ breast cancer seen in these CHS are not widely published.

This analysis describes the systemic treatment utilization across a sample of CHS.

METHODS

A retrospective analysis was performed utilizing the Syapse Learning Health Network™ (LHN), an electronic medical record (EMR) derived database that collects cancer care data from multiple care settings within CHS. Data utilized for this analysis included both structured data (set EMR fields like sex and birth date) and unstructured data (e.g. physician notes) validated by Syapse's Certified Tumor Registrars and then descriptively summarized.

Syapse Learning Health Network™

33 states

450+ hospitals

1,900+ community employed oncologists

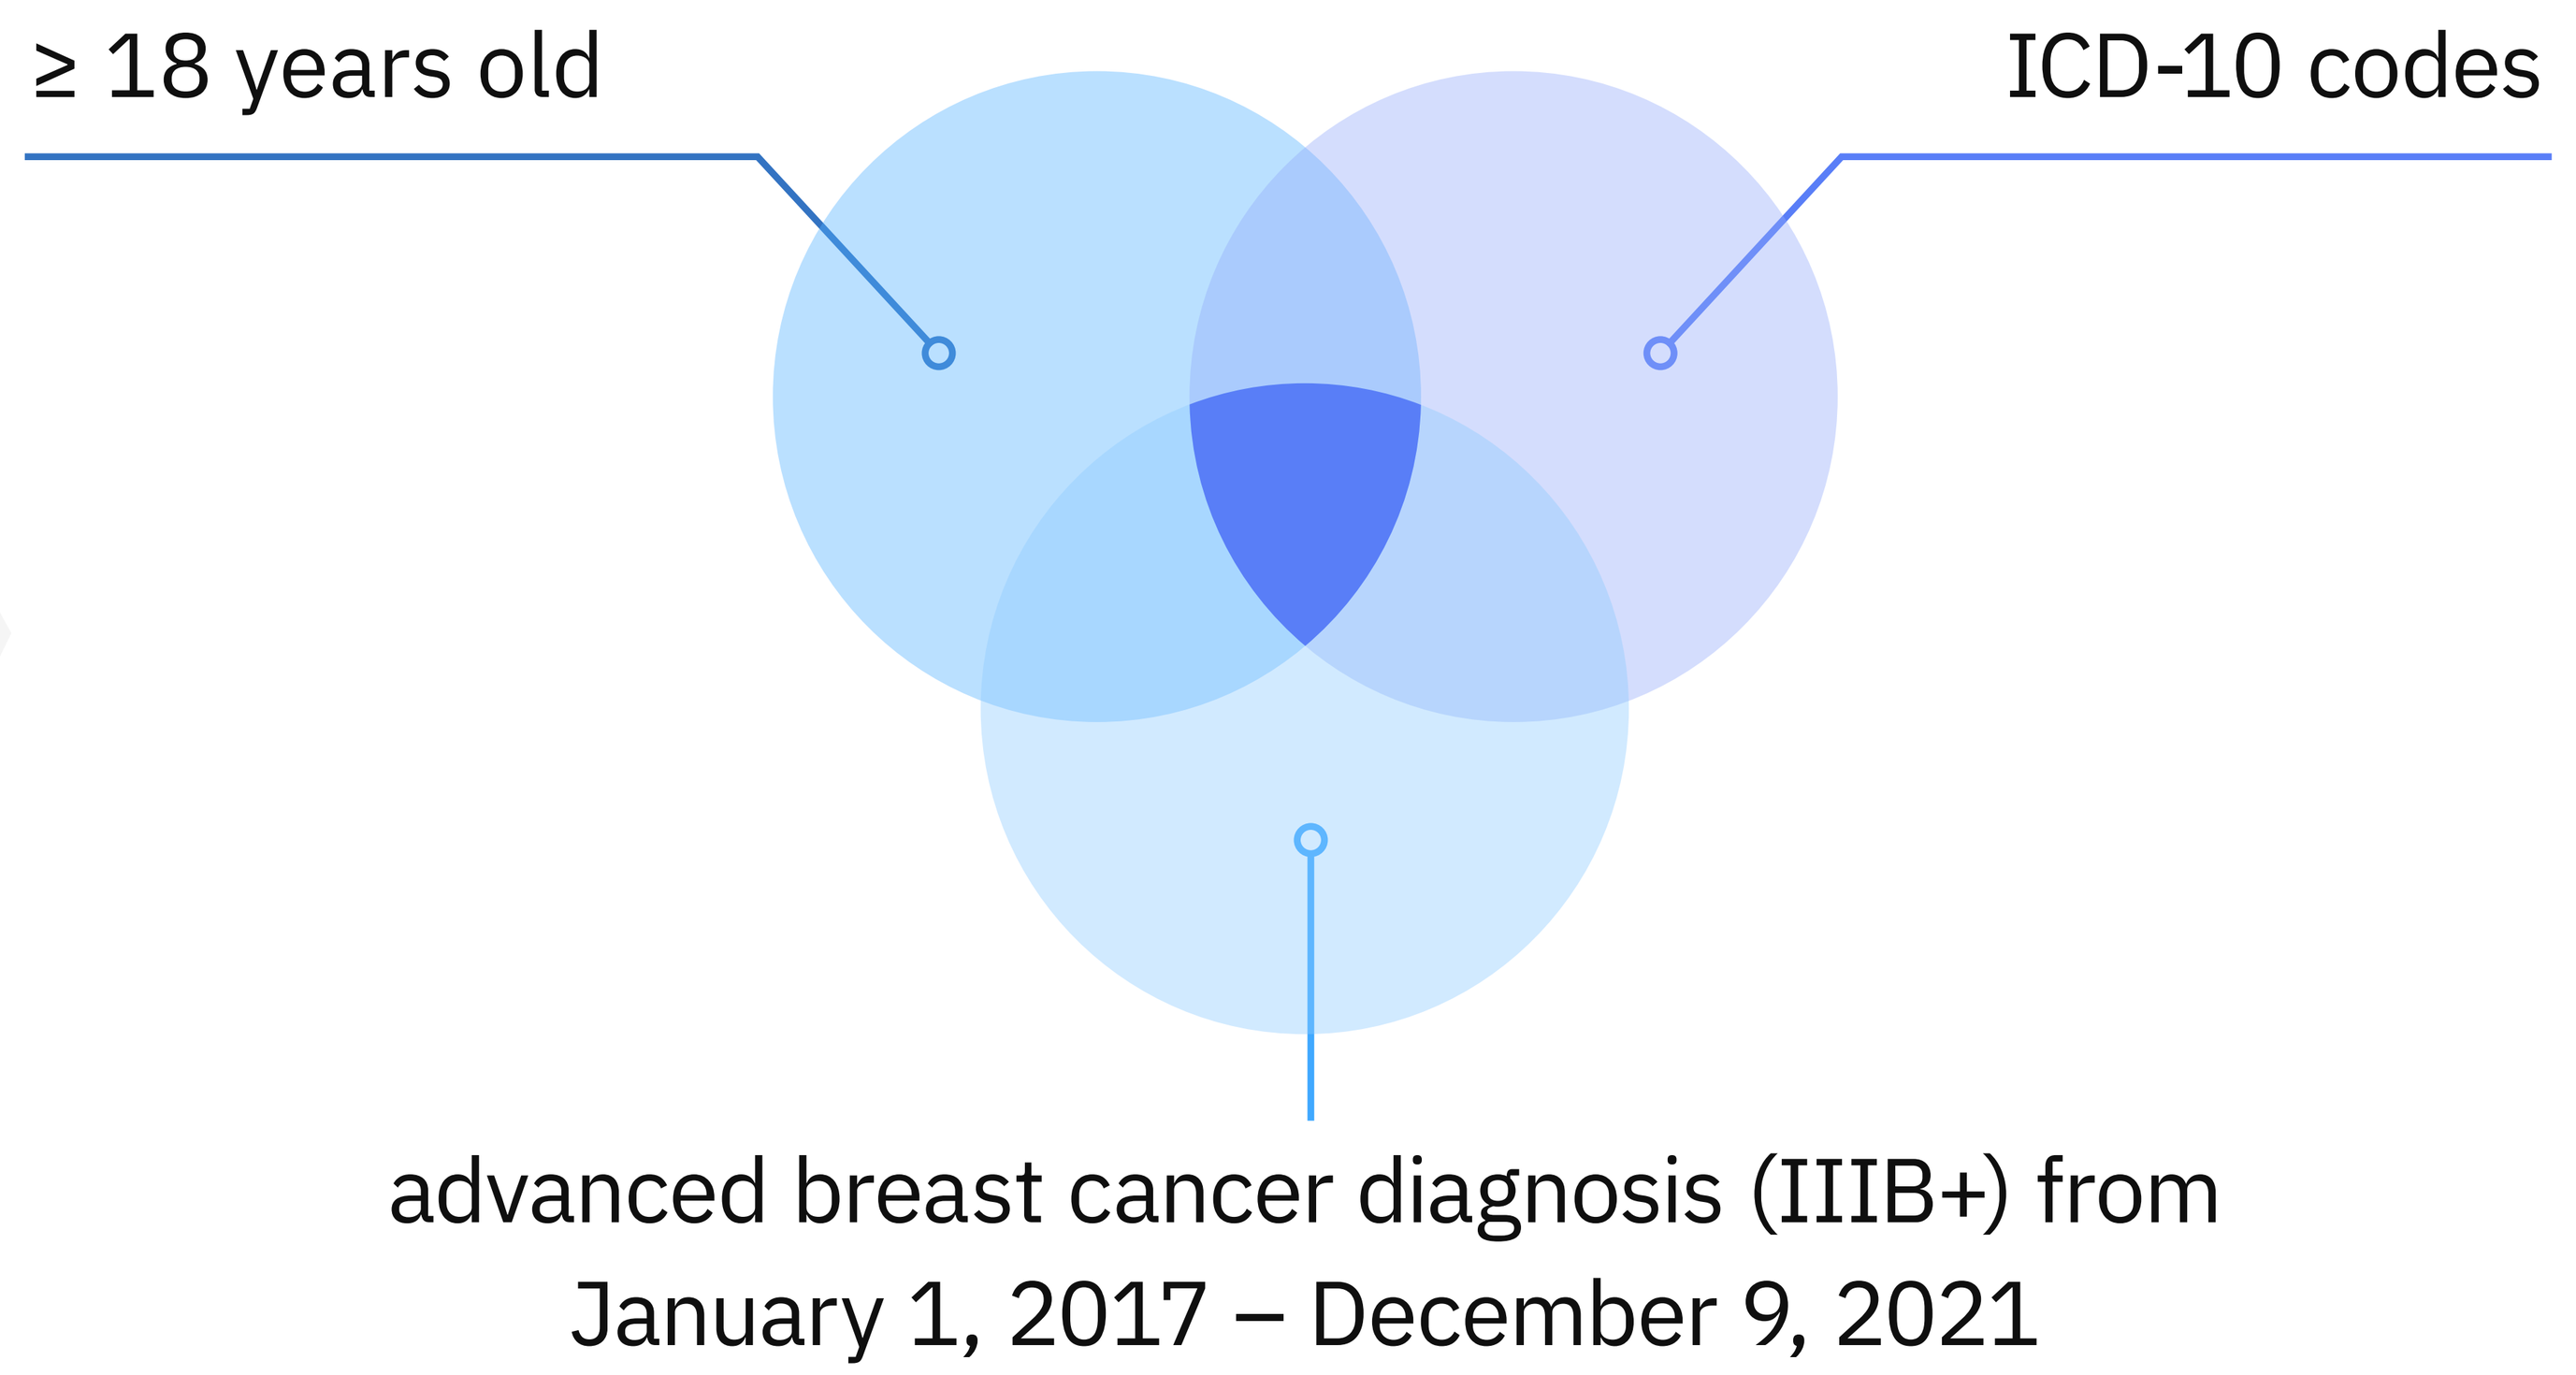
≥ 18 years old
ICD-10 codes
advanced breast cancer diagnosis (IIIB+) from January 1, 2017 – December 9, 2021

HER2+ BREAST CANCER RESULTS

692

patients aged 18+ with advanced HER2+ breast cancer (stage IIIB+)

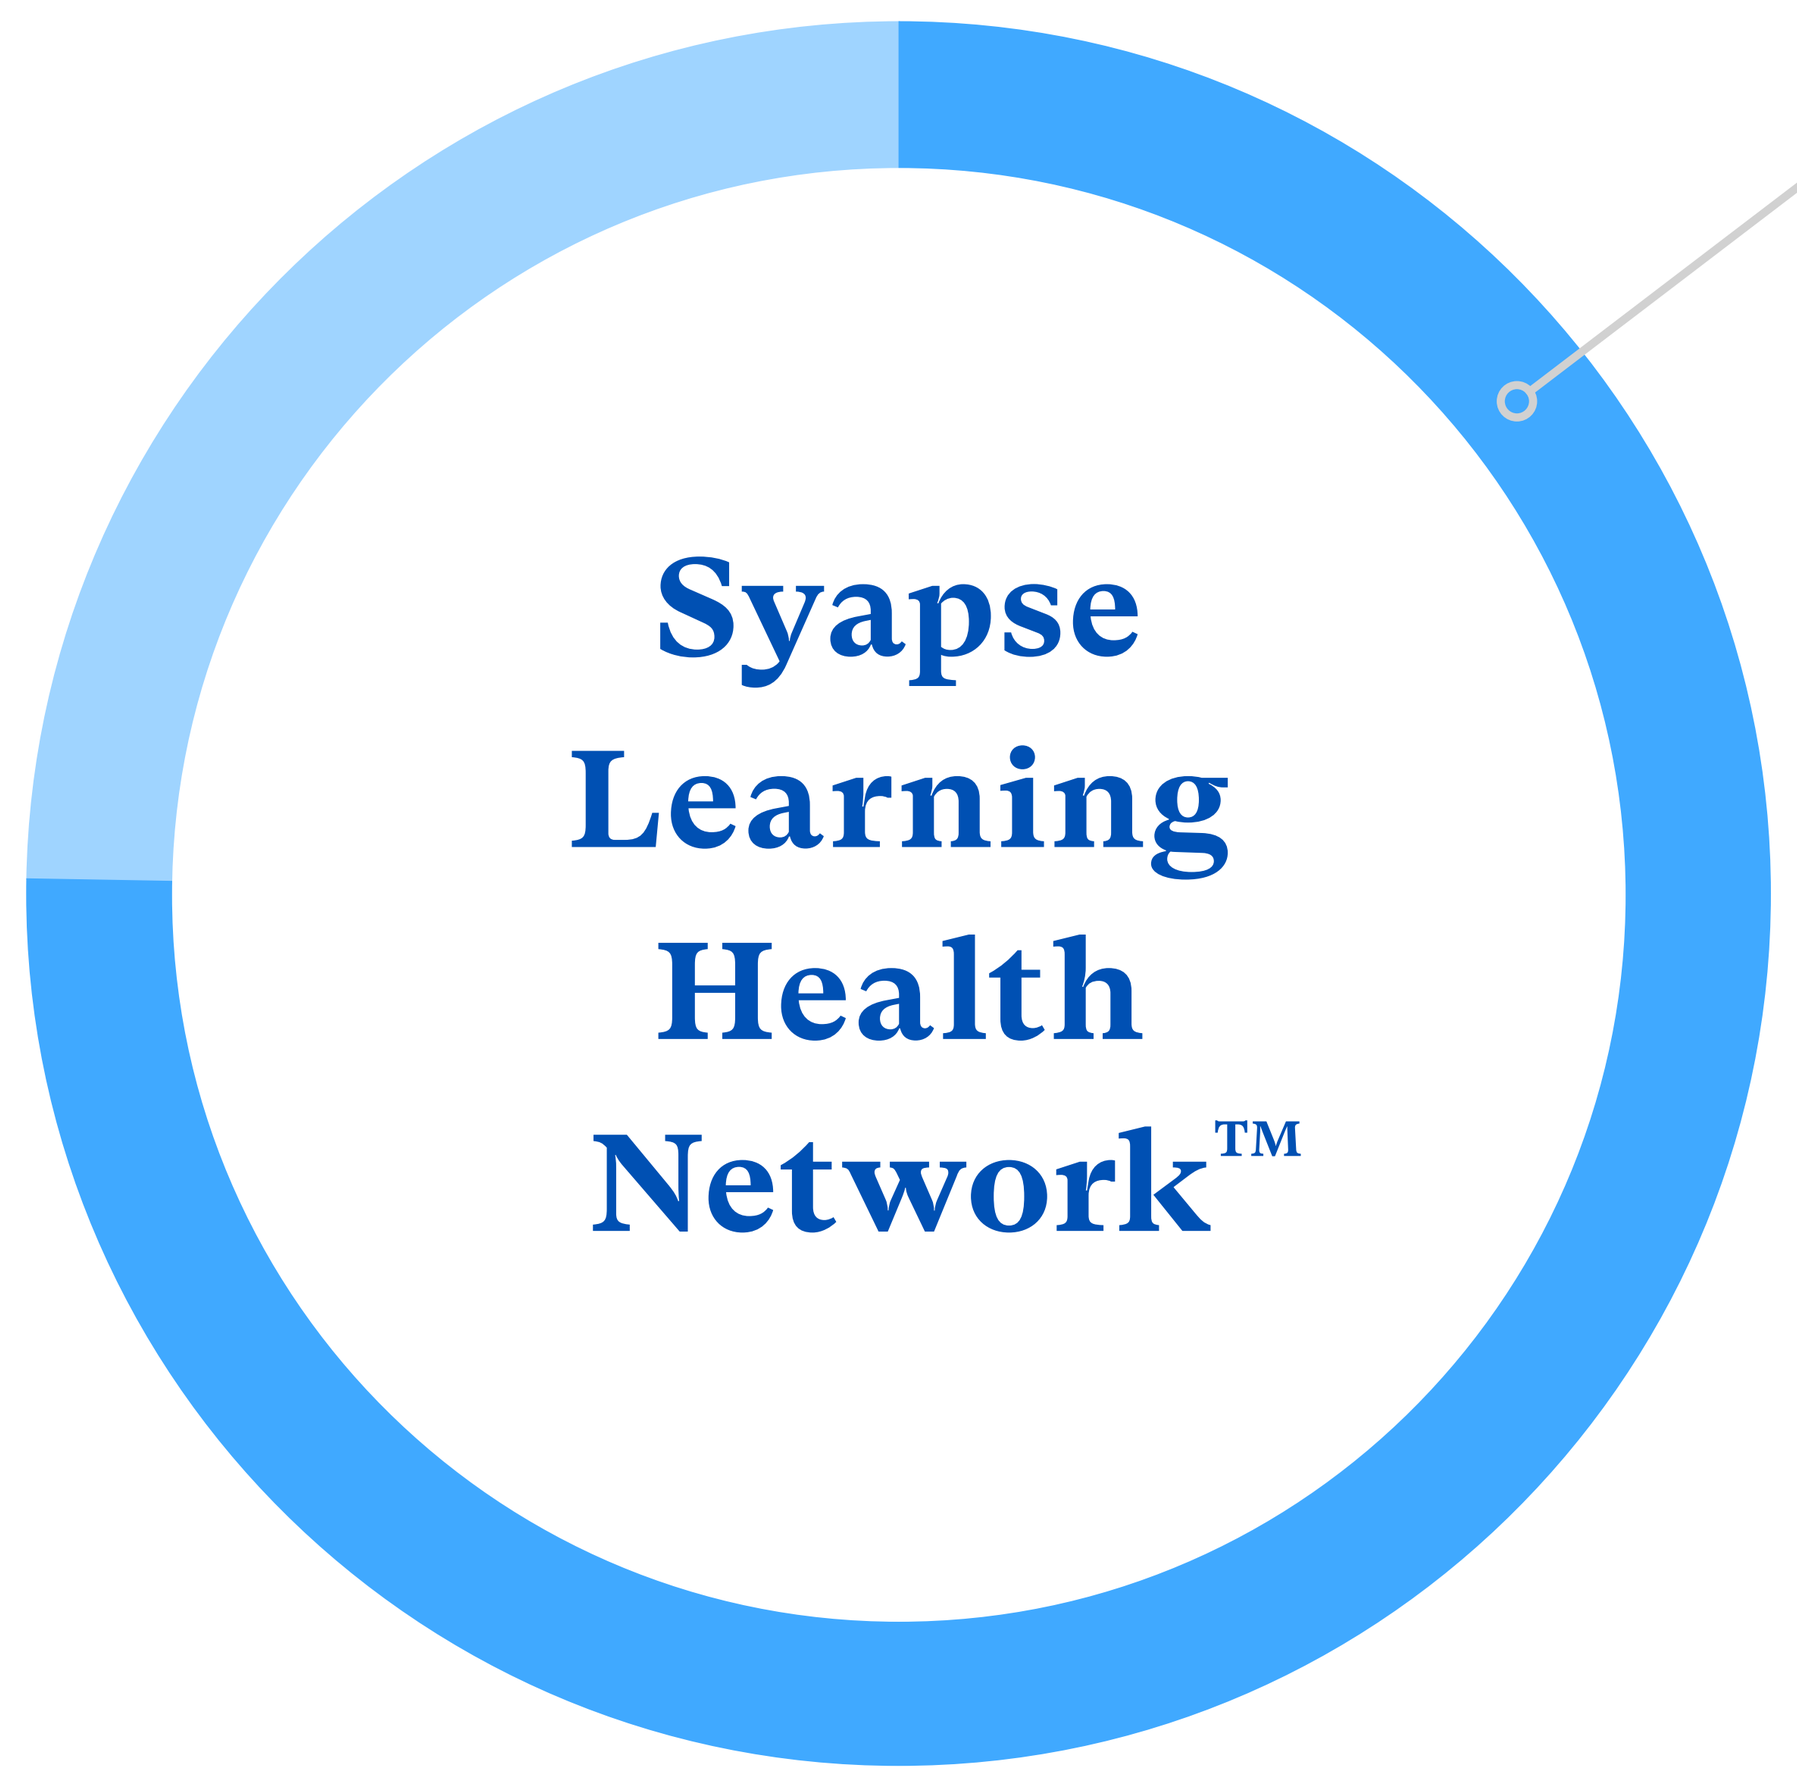
Syapse Learning Health Network™

TREATMENT

521

patients received antineoplastic systemic treatment in 1st line (1L) setting

80%	79%	18%
CHEMOTHERAPY	TARGETED THERAPY	HORMONE THERAPY

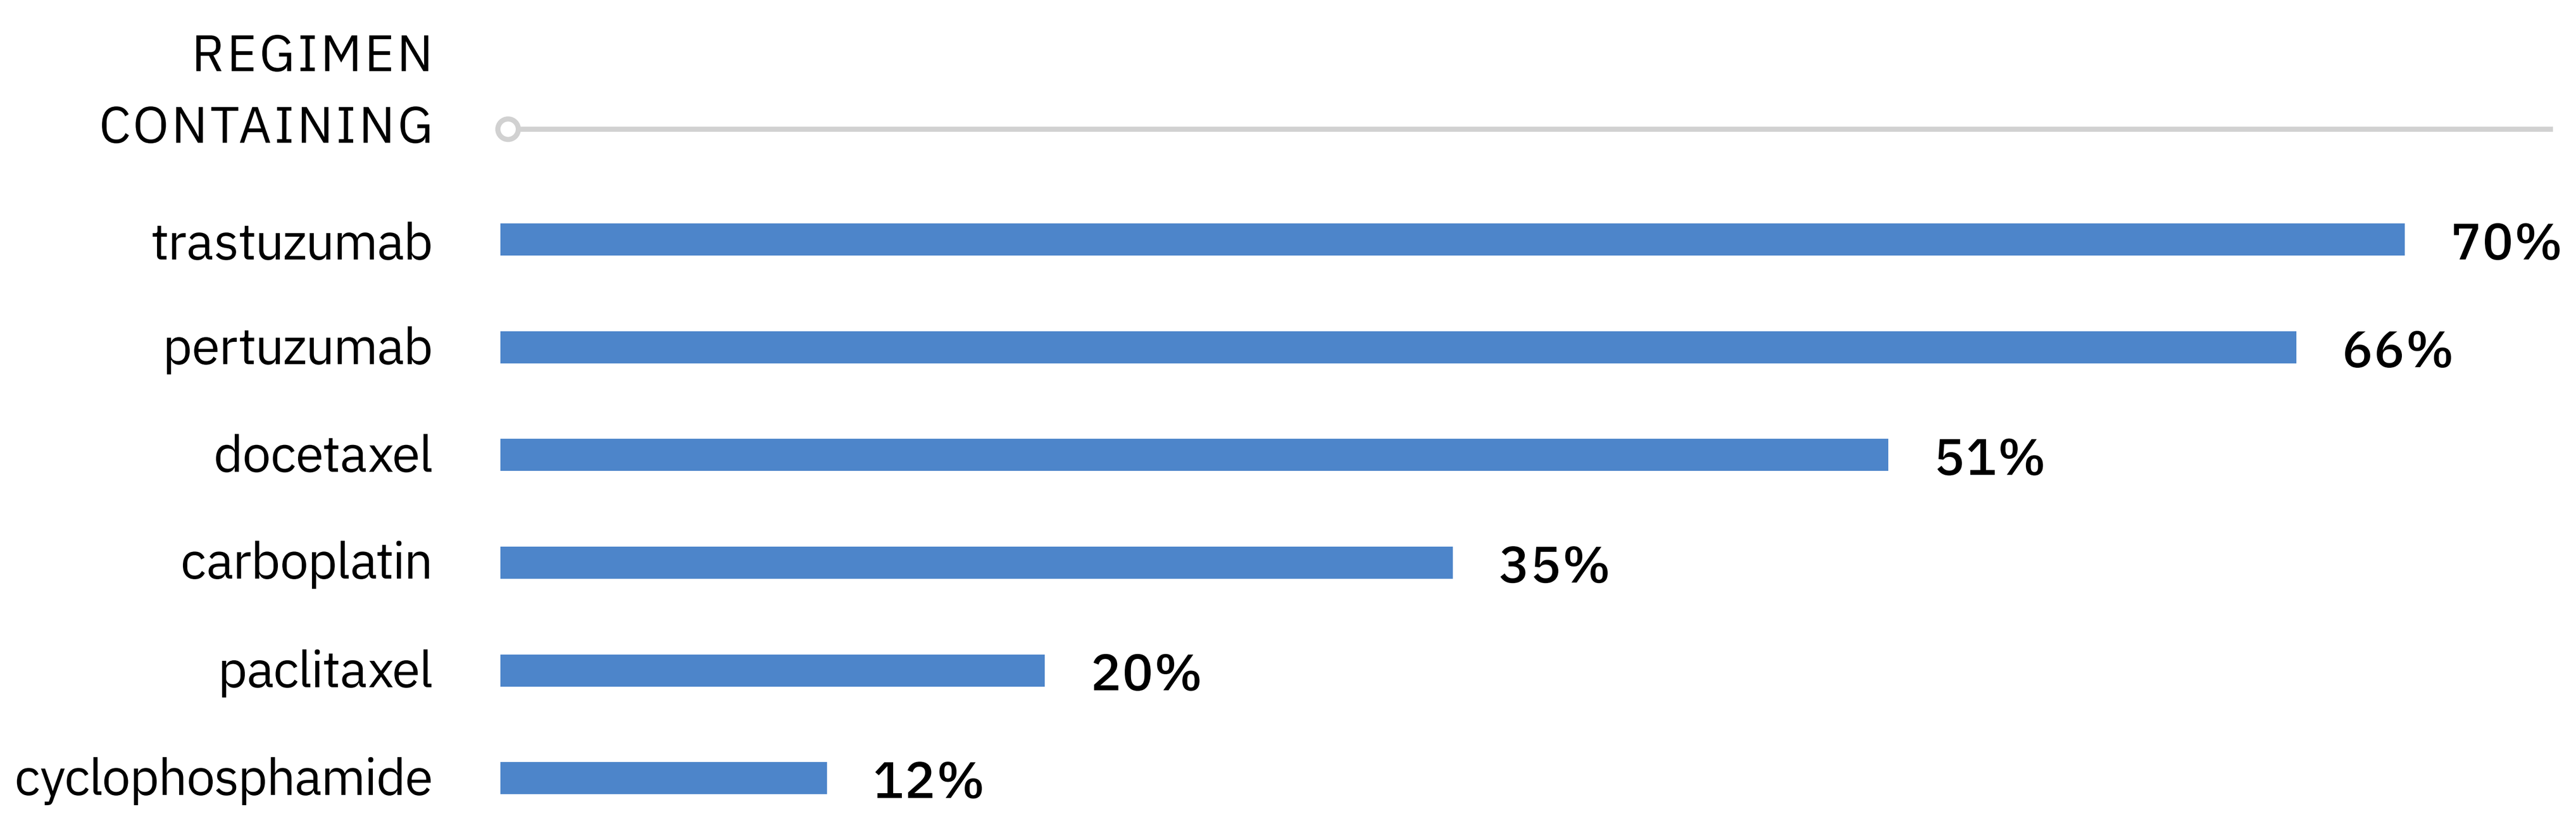
REGIMEN CONTAINING
trastuzumab 70%
pertuzumab 66%
docetaxel 51%
carboplatin 35%
paclitaxel 20%
cyclophosphamide 12%

CONCLUSIONS

Consistent with management guidelines, HER2+ patients with advanced breast cancer treated within CHS were most likely to receive 1L chemotherapy and HER2 targeted therapies, either trastuzumab or pertuzumab. Further analysis is necessary to better understand management decisions for this group of patients cared for within CHS.