Comparison of Healthcare Resource Utilization and Costs in Patients With and Without Bullous Pemphigoid: A Retrospective Analysis of US Health Insurance Claims Data

Xiao Xu[1], Catherine Datto[1], Justin Kwiatek[1], Juliana Meyers[2], Sean Candrilli[2], Daniel Mines[2], Heide Stirnadel-Farrant[3]
[1]AstraZeneca, Gaithersburg, MD, USA; [2]RTI Health Solutions, Research Triangle Park, NC, USA; [3]AstraZeneca, Cambridge, UK.

Presented at ISPOR, May 15-18, 2022, Washington, DC, USA

Please scan for an electronic version of this poster

Background

- Bullous pemphigoid (BP) is a rare chronic autoimmune disease mainly affecting the elderly with a global incidence rate of 34.2 per million person-years.[1]
 - Patients typically present with pruritis or pain, along with tense blisters and skin erosions, with periodic exacerbations.[2]
- High-potency topical steroids are typically first-line therapy for BP, followed by systemic glucocorticoids.
- As BP is largely a disease of the elderly, patients often have a range of comorbidities and also appear to be at an increased risk for mortality as well as viral and bacterial infection.[3,4] Chronic and high-dose steroid use are also associated with adverse events.[5]
- Despite this, limited real-world data exist regarding the healthcare resource utilization (HCRU) and costs among patients with BP.

Objective

- To evaluate all-cause and BP-related HCRU and costs among patients with BP, including the incremental burden of disease, compared with a population of patients without BP.

Methods

Study design

- Administrative claims (2015-2019) from the IBM MarketScan US Commercial and Medicare databases were retrospectively assessed.
- Patients with BP (BP cohort) were matched with patients without BP (non-BP cohort), based on demographic and clinical characteristics.
- Patients were followed until either health plan disenrollment or end of study database, whichever was the earliest.

BP cohort

- ≥18 years on date of first observed BP diagnosis (i.e. index date).
- ≥1 pharmacy claim for BP therapy within ±30 days of the index date.
- No diagnosis of pemphigus at any point.
- ≥6 months continuous health plan enrolment pre-index, no diagnosis of BP during this time.

Non-BP cohort

- Assigned index date of matched BP case.
- ≥6 months continuous health plan enrolment pre-index date and a follow-up period equivalent to their matched BP case.
- No diagnosis of BP or pemphigus at any point after first medical claim date.

Figure 1. Study schema

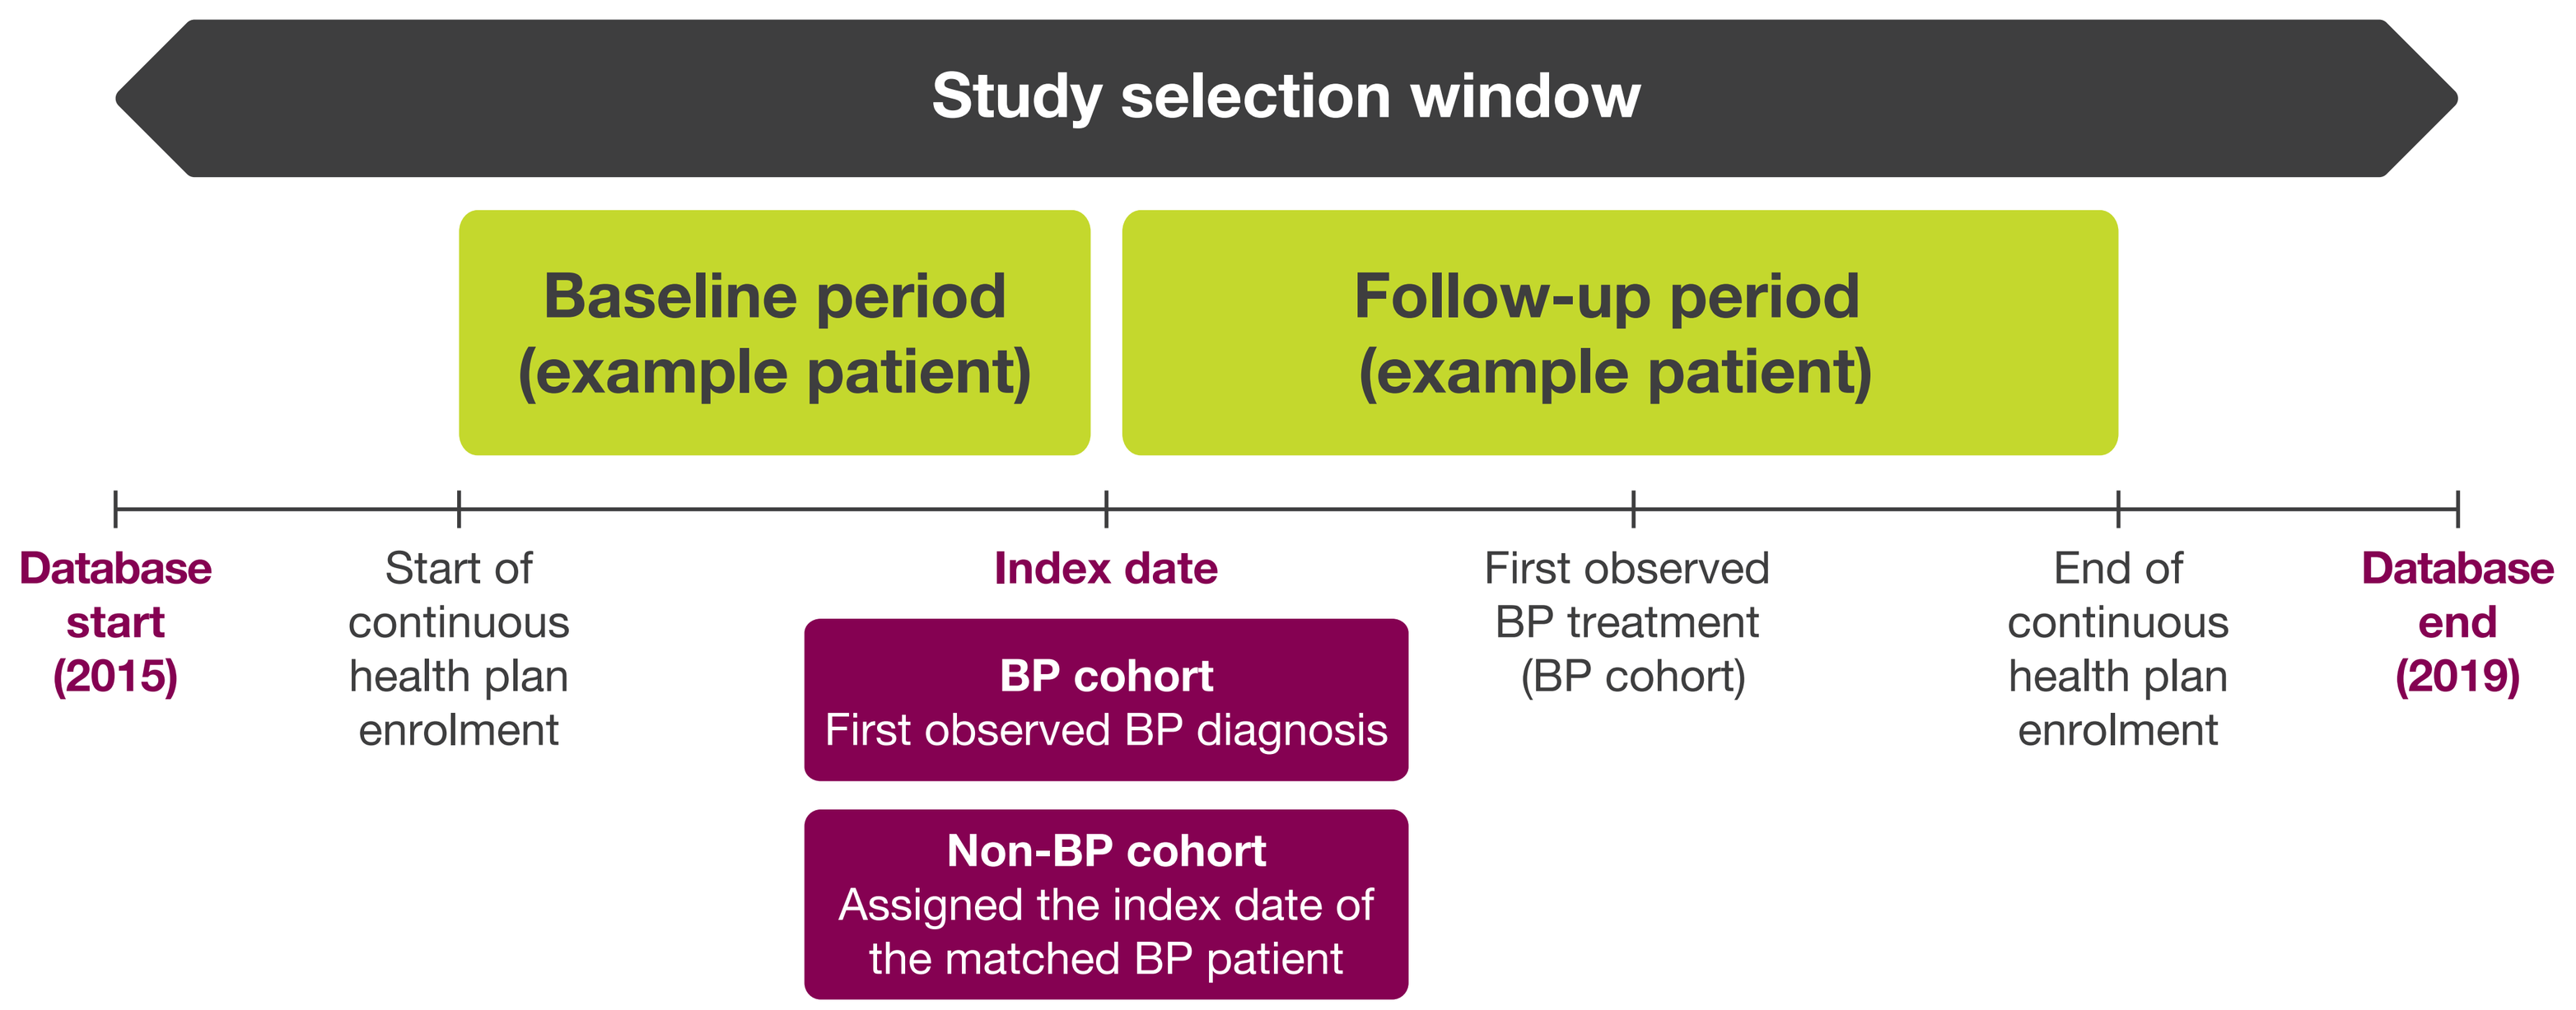
Study selection window
Baseline period (example patient)
Follow-up period (example patient)
Database start (2015)
Start of continuous health plan enrolment
Index date
BP cohort First observed BP diagnosis
Non-BP cohort Assigned the index date of the matched BP patient
First observed BP treatment (BP cohort)
End of continuous health plan enrolment
Database end (2019)

Outcomes

All-cause HCRU were assessed by service setting:

- Hospitalization*
- Ambulatory surgical center visits
- Emergency department (ED) visits
- Urgent care visits
- Outpatient visits†
- Physician office visits
- Home visits
- Other outpatient and ancillary care
- Telemedicine consultation
- Pharmacy prescriptions

*Patients with at least one inpatient hospital admission; †Patients with at least one outpatient hospital visit.

- All-cause HCRU costs were reported as *per-patient-per-month* (**PPPM**) and annualized, and adjusted to 2020 US dollars using the medical care component of the US Consumer Price index.[6]

Statistical methods

- The BP and non-BP cohorts were matched at a ratio of up to 1:5 using direct covariate matching based on demographic and clinical characteristics (year of birth, sex, geographic region, baseline Charlson Comorbidity Index [CCI] score, months of follow-up).
 - The CCI is a weighted assessment tool used to predict long-term mortality in patients with multiple comorbidities. Each comorbidity is assigned a score from 1 to 6, with higher scores indicating a stronger association with mortality or higher resource use.[7]
- Descriptive analyses of patient demographics, clinical characteristics, HCRU and associated costs were reported.
- Multivariable regression analyses were conducted to estimate adjusted all-cause costs among patients in the BP and matched non-BP cohorts.
- All analyses were conducted using SAS Studio (Cary, NC, USA).

Results

Patient demographics and clinical characteristics at the index date

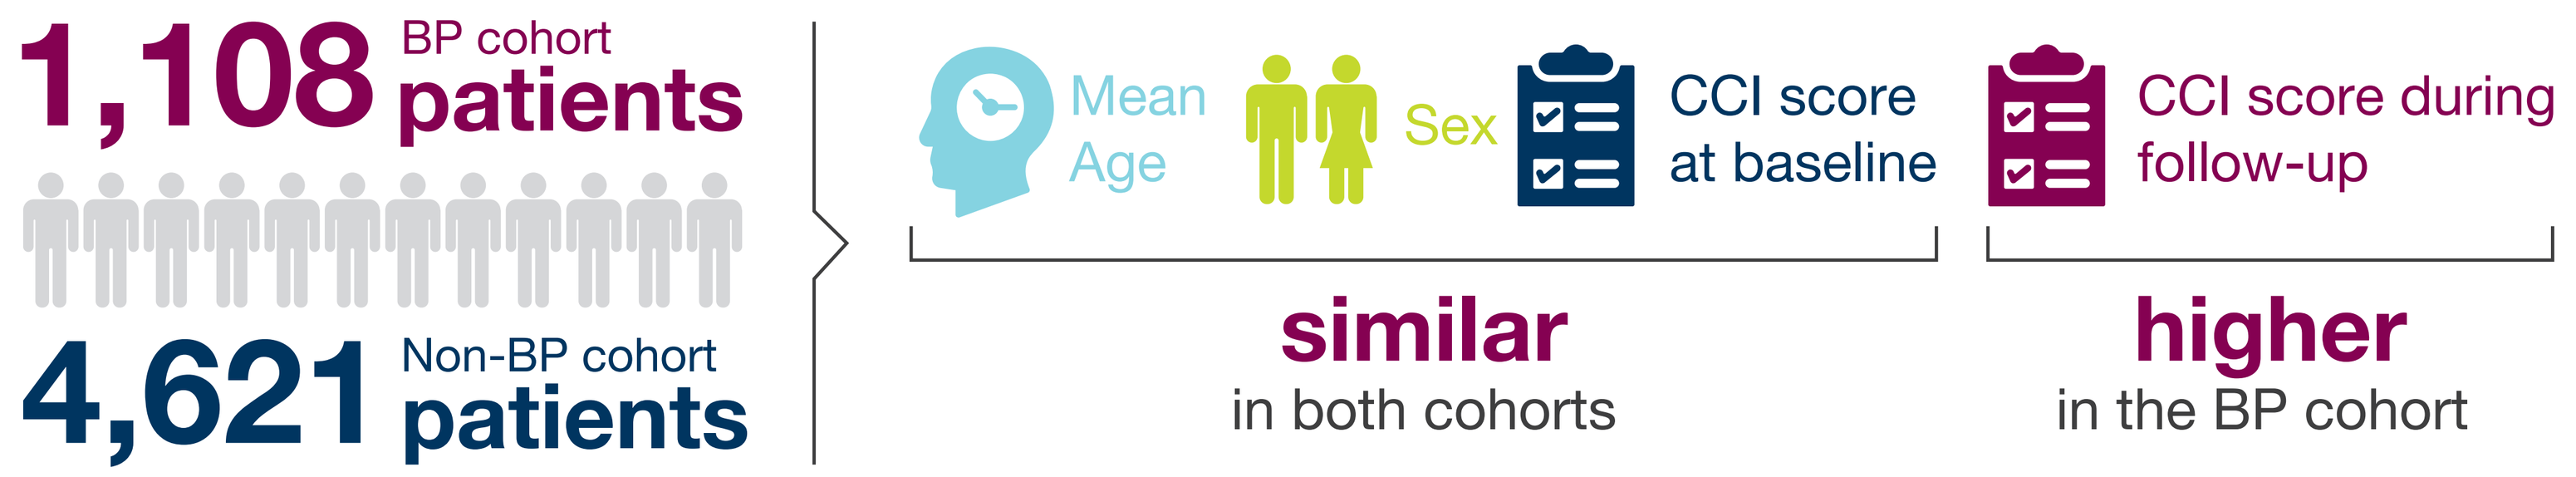
1,108 BP cohort patients
4,621 Non-BP cohort patients
Mean Age
Sex
CCI score at baseline
CCI score during follow-up
similar
in both cohorts
higher
in the BP cohort

Table 1. Patient demographics and clinical characteristics

Characteristic	BP cohort (N=1,108)	Non-BP cohort (N=4,621)
Mean (SD) age at index, years	73.6 (14.9)	71.1 (14.7)
Female, %	53.4	54.3
Mean (SD) CCI score at baseline	3.3 (2.7)	2.8 (2.4)
Mean (SD) CCI score in the follow-up period	5.0 (4.9)	3.7 (3.0)
Payer type, %		
Commercial	32.0	38.3
Medicare	70.9	61.7

BP, bullous pemphigoid; CCI, Charlson Comorbidity Index; SD, standard deviation.

PPPM all-cause HCRU

- The prevalence of hospitalization for BP patients was substantially greater in the follow-up period compared to the baseline period (44% vs 15%). The lengths of stay per hospitalization were similar (6.0 vs 6.4 days).
- The non-BP cohort had similar HCRU in the baseline and follow-up periods (**Table 2**).

Table 2. PPPM all-cause HCRU

Mean HCRU PPPM	Baseline period		Follow-up period	
	BP cohort	Non-BP cohort	BP cohort	Non-BP cohort
Hospitalization	0.18	0.10	0.77	0.25
Ambulatory surgical center claims	0.06	0.07	0.05	0.05
ED claims	0.24	0.11	0.23	0.12
Urgent care claims	0.02	0.01	0.02	0.02
Outpatient claims	2.58	1.56	3.03	1.57
Physician office claims	3.31	2.50	3.68	2.32
Home claims	0.47	0.24	0.84	0.24
Other outpatient care claims	2.02	1.01	3.15	1.02

BP, bullous pemphigoid; ED, emergency department; HCRU, healthcare resource utilization; PPPM, per-patient-per-month.

PPPM all-cause total healthcare costs

- In the baseline period, regardless of cohort, outpatient visits were the main HCRU cost component, followed by hospitalization (**Figure 2**).
 - PPPM all-cause total healthcare costs in the BP cohort were almost double those in the non-BP cohort ($2,856 vs $1,566); annualized difference was $15,480.
- In the follow-up period, hospitalization was the main cost component for both BP and non-BP patients, followed by outpatient visits (**Figure 2**).
 - PPPM all-cause total healthcare costs increased 13% between baseline and follow-up for the BP cohort, mainly driven by increased hospitalization costs, but decreased 14% for the Non-BP cohort.
- PPPM all-cause total healthcare costs in the follow-up period were almost 2.5 times greater for the BP cohort versus the non-BP cohort (**Figure 2**).
 - The difference, when annualized, amounted to a $22,332 per year higher cost for BP patients compared with matched non-BP patients.

Figure 2. PPPM all-cause total healthcare costs

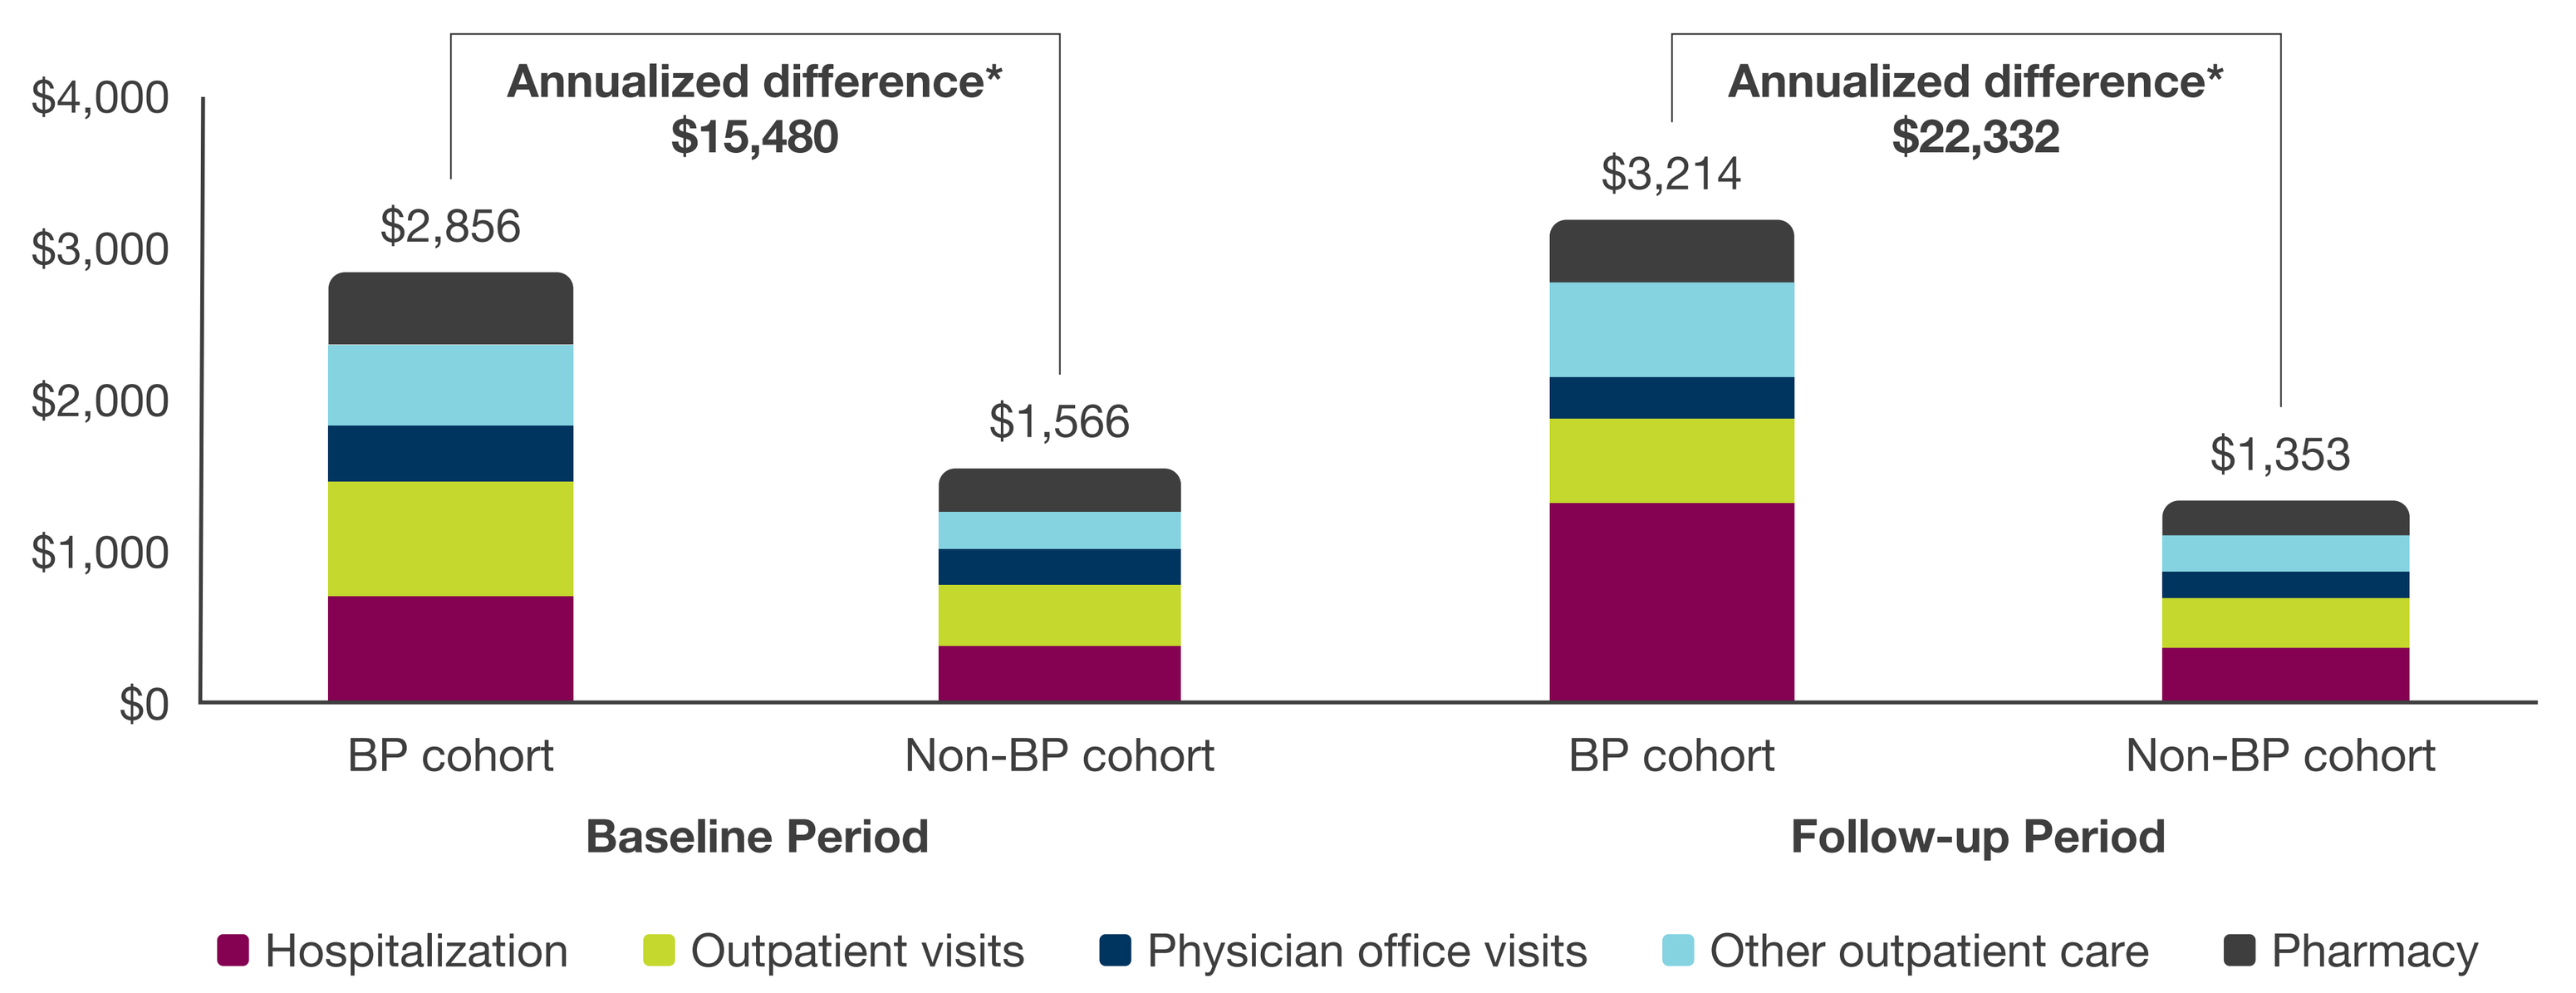
$4,000
$3,000
$2,000
$1,000
$0
Annualized difference* $15,480
Annualized difference* $22,332
$2,856
$1,566
$3,214
$1,353
BP cohort
Non-BP cohort
BP cohort
Non-BP cohort
Baseline Period
Follow-up Period
Hospitalization
Outpatient visits
Physician office visits
Other outpatient care
Pharmacy

*PPPM all-cause total healthcare costs were annualized by multiplying by 12.
Other outpatient care includes ambulatory surgical center visits, emergency department visits, urgent care visits, home visits, telemedicine consultations, and other outpatient and ancillary care.
BP, bullous pemphigoid; PPPM, per-patient-per-month.

Adjusted PPPM all-cause total healthcare costs

- PPPM costs in the baseline and follow-up periods were slightly higher for both cohorts after adjusting for baseline age, gender, payer type, region, year of index date and CCI score, but the magnitude of differences between cohorts were similar to unadjusted values (**Figure 3**).

Figure 3. Adjusted PPPM all-cause total healthcare costs

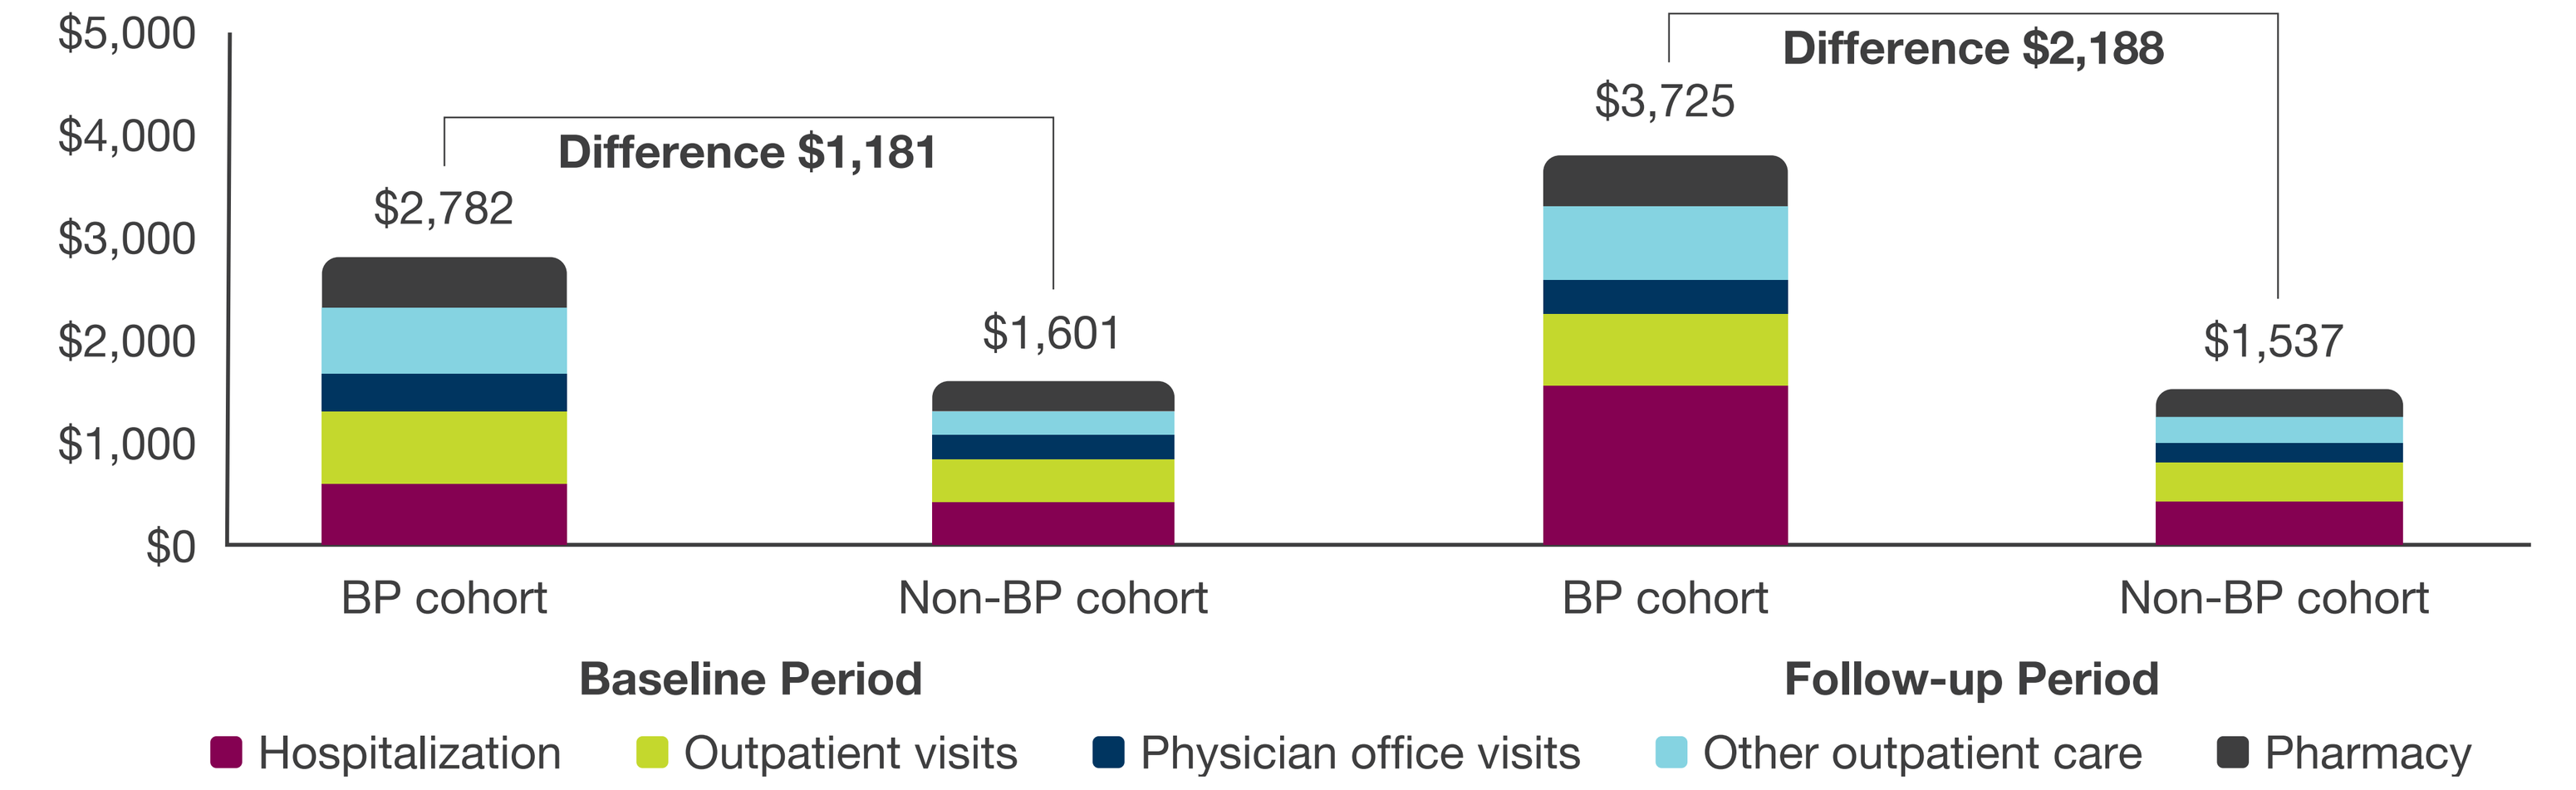
$5,000
$4,000
$3,000
$2,000
$1,000
$0
Difference $1,181
Difference $2,188
$2,782
$1,601
$3,725
$1,537
BP cohort
Non-BP cohort
BP cohort
Non-BP cohort
Baseline Period
Follow-up Period
Hospitalization
Outpatient visits
Physician office visits
Other outpatient care
Pharmacy

Other outpatient care includes ambulatory surgical center visits, emergency department visits, urgent care visits, home visits, and other outpatient or ambulatory care.
BP, bullous pemphigoid; CCI, Charlson Comorbidity Index; PPPM, per-patient-per-month.

Subgroup analysis

- Among the subset of patients who had 12-months of baseline and follow-up data, the difference between BP (n=448) and non-BP patients (n=1,946) was not as pronounced ($17,092) as annualized PPPM calculations (**Figure 4**).

Figure 4. All-cause total healthcare costs in the 12-month follow-up period

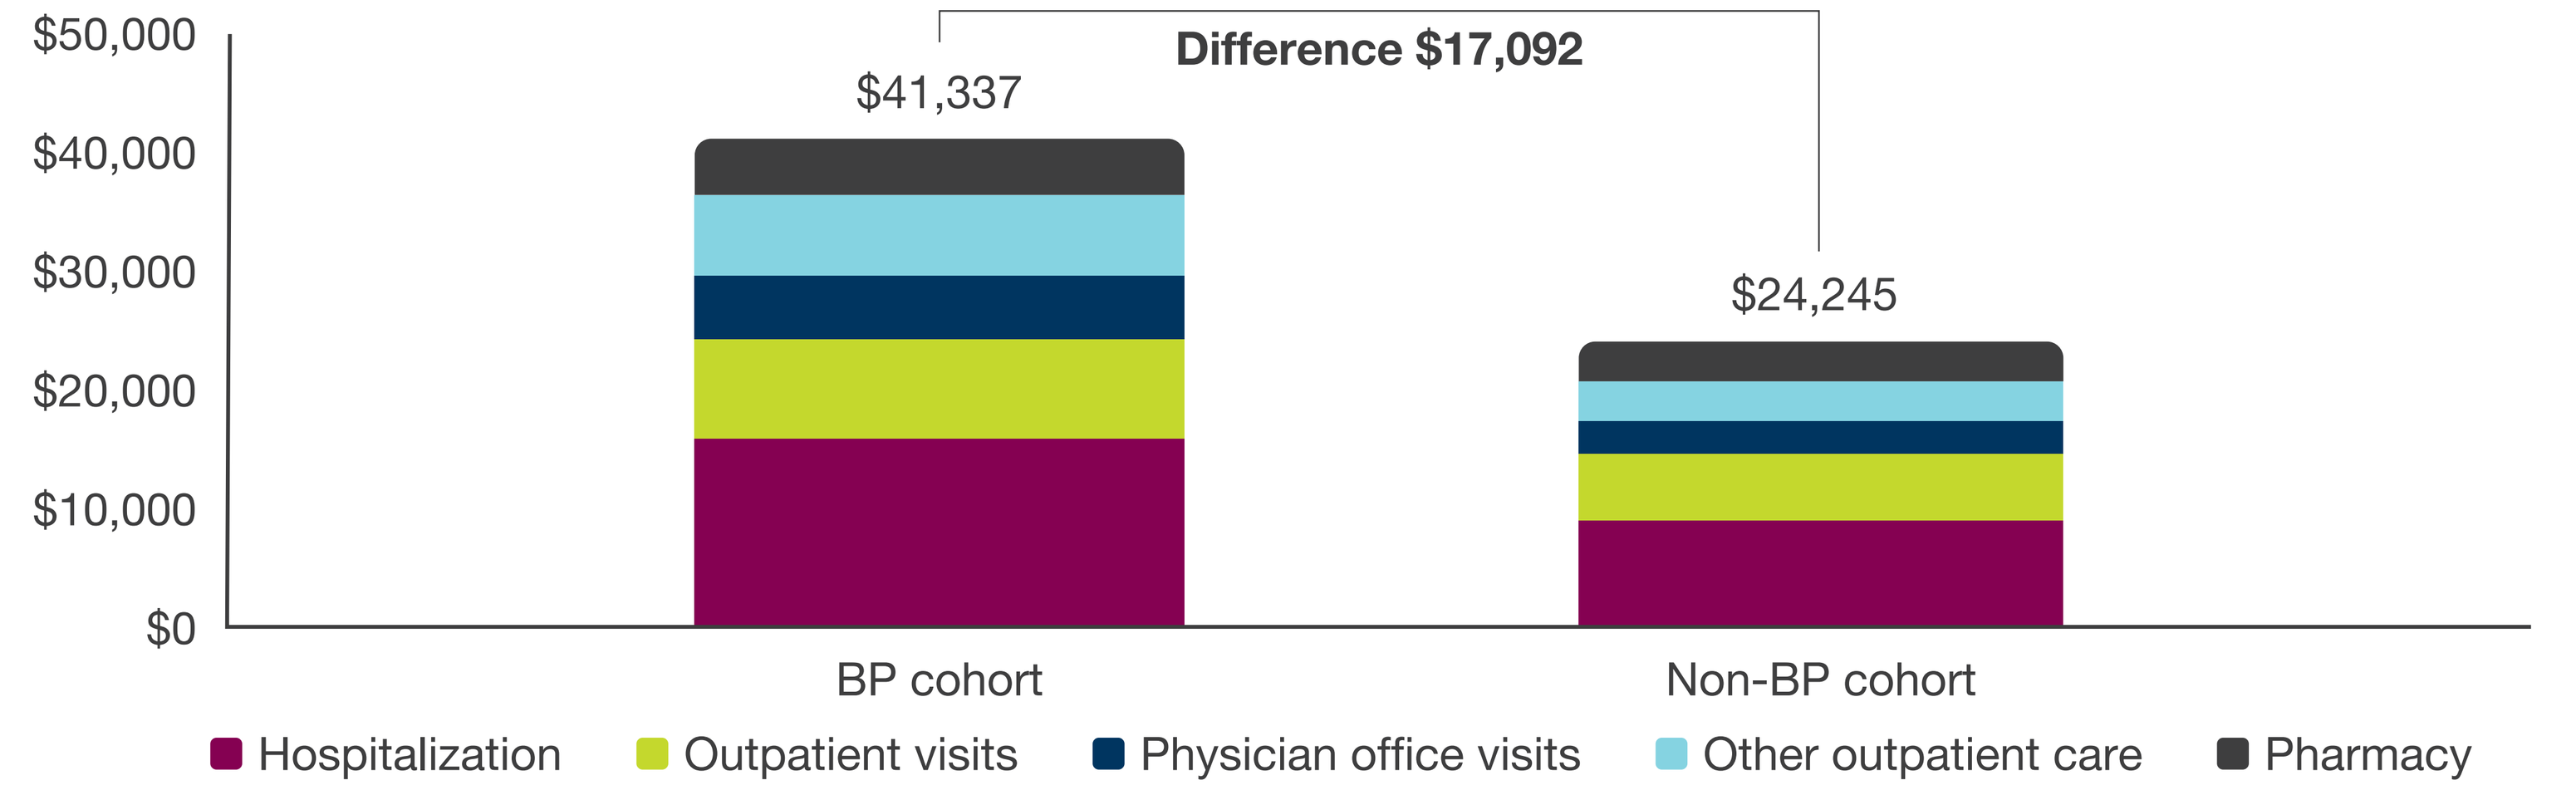
$50,000
$40,000
$30,000
$20,000
$10,000
$0
Difference $17,092
$41,337
$24,245
BP cohort
Non-BP cohort
Hospitalization
Outpatient visits
Physician office visits
Other outpatient care
Pharmacy

Other outpatient care includes ambulatory surgical center visits, emergency department visits, urgent care visits, home visits, and other outpatient and ancilliary care.
BP, bullous pemphigoid.

Limitations

- Diagnoses were based on billing codes which are subject to inaccuracies, such that some patients may not have genuinely had BP. However, the requirement of two diagnoses of BP at least 30 days apart, plus receipt of a BP-related medication, should have substantially limited patients without BP being included in the study sample.
- Claims data do not include clinical measurements that may affect the management of BP, such as functional status, mucous membrane involvement, and lifestyle factors.
- The MarketScan data are based on employment-related insurance claims, so some information may be missing from before and after employment for non-retired patients.
- As BP is a relapsing remitting disease, the date of disease onset may not be accurate.
- No information on reason for health plan disenrollment (e.g. death) is available in the database.

Conclusions

- BP patients had higher CCI post-diagnosis and substantially higher PPPM HCRU than matched non-BP patients, resulting in higher mean total PPPM medical encounter costs.
- The largest component of cost (42%) for BP patients in the follow-up period was hospitalization, with 44% of patients having ≥1 hospitalization, with an average length of stay of 6.4 days.
- A greater difference was seen between BP and non-BP patients with annualized data from PPPM calculations ($22,332) than with 12-month follow-up data ($17,092), suggesting that patients may have higher HCRU at diagnosis or potentially higher end of life costs, which lowered later in the year.
- This analysis represents the first real-world estimates of HCRU and costs among patients with BP. Our results indicate that more effective and targeted treatments are needed for BP without the potential adverse events associated with oral or systemic glucocorticoids.

References
1. Persson MSM, et al. *Br J Dermatol* 2022;186(3):414-425; 2. Wertenteil S, et al. *J Am Acad Dermatol* 2019;80(3):655-659; 3. Lee S, et al. *Arch Dermatol Res* 2021;313(5):327-332; 4. Langan SM, et al. *BMJ* 2008;337:a180; 5. Liu D, et al. *All Asth Clin Immun* 2013;9(30); 6. U.S. Bureau of Labor Statistics. Consumer Price Index; https://www.bls.gov/cpi/; 7. Charlson ME, et al. *J Chronic Dis* 1987;40(5):373-383.

Acknowledgments
We would like to thank Stefan Courtney of inScience Communications, Springer Healthcare, for providing medical writing support, which was funded by AstraZeneca in accordance with Good Publication Practice (GPP3) guidelines (http://www.ismpp.org/gpp3).

Disclosures
XX, CD and HS-F are full-time employees of AstraZeneca. JK is a full-time employee of, and owns stock in, AstraZeneca. JM, SC and DM are employees of RTI Health Solutions, which received funding from AstraZeneca for the conduct of this study.