Long-term Health Outcomes in Patients With Schizophrenia Treated With the Long-Acting Injectable Antipsychotic Aripiprazole Lauroxil for 1 Year

Sabina Gasper, Steve Dingman, Meihua Wang, Brittany D. Roy

Alkermes, Inc., Waltham, MA, USA

INTRODUCTION

- Patients with schizophrenia experience disability across multiple functional domains, with poor social and occupational outcomes[1]
- Symptomatic remission is associated with improvement in functional outcomes[2,3]; however, at least 50% of patients in symptomatic remission fail to achieve adequate functioning or functional recovery[2,4]
 - Functional recovery is nonetheless considered an achievable goal for patients with schizophrenia; consequently, assessment of patient functioning is essential in clinical research[5]
- The long-acting injectable (LAI) antipsychotic aripiprazole lauroxil (AL), at a dose of 441 or 882 mg every 4 weeks (q4wk), significantly reduced Positive and Negative Syndrome Scale[6] (PANSS) total score from baseline versus placebo in patients with schizophrenia in a pivotal 12-week efficacy study[7]
- The pivotal trial was followed by a phase 3, 52-week, open-label extension (OLE) study (NCT01626456) conducted to assess long-term safety and durability of response to AL in patients with schizophrenia[8,9]
 - As previously reported,[8] treatment with AL provided durable efficacy over 52 weeks; mean (SD) change in PANSS total score from baseline to 1 year was –8.1 (9.77)
 - Patient- and clinician-rated functional outcomes were also assessed in the OLE study using the EuroQol Group Health Outcome Measure-five level[10] (EQ-5D-5L) and the Personal and Social Performance (PSP) scale,[11] respectively

OBJECTIVES

- To assess health outcomes using the EQ-5D-5L and PSP scale in patients who remained on AL for up to 1 year

METHODS

Study Design and Assessments

- This was a prespecified analysis from a phase 3 long-term safety study of 2 fixed doses of AL (441 or 882 mg q4wk),[9] which enrolled patients who
 - **either** had completed the pivotal phase 3, 12-week, randomized, double-blind, placebo-controlled AL trial[6]
 - **or** were enrolled de novo while receiving a stable dose of oral antipsychotic medication
- All patients were assigned to receive 52 weeks of open-label treatment with AL
 - Dose level was blinded for those who completed the lead-in study
 - Patients assigned to receive the 441- or 882-mg AL treatment in the lead-in study continued on their previously assigned AL dose
 - Patients previously assigned to receive placebo started AL 441 or 882 mg q4wk based on their placebo assignment (matched to either a low- or high-volume placebo injection, respectively)
 - Patients who enrolled de novo were assigned to receive AL 882 mg q4wk
- Patients received their first intramuscular AL injection on study day 1, followed by an injection every 4 weeks thereafter (last injection, week 48)

Functional Outcomes Assessments

- EQ-5D-5L
 - Patients scored 5 health dimensions: mobility, self-care, usual activities, pain/discomfort, and anxiety/depression
 - Each dimension included a 5-level scale: no problems, slight problems, moderate problems, severe problems, and extreme problems
- PSP scale[11]
 - Clinicians rated patient functioning across 4 domains: socially useful activities (eg, work and study), personal and social relationships, self-care, and disturbing and aggressive behaviors
 - PSP total score interpretation: 1–10, lack of autonomy; 11–40, severe difficulties; 41–60, marked difficulties; 61–70, manifest but not marked difficulties; 71–80, mild difficulties; 81–100, good to excellent functioning

Study Population

- Adult outpatients aged ≥18 to ≤70 years at screening with a diagnosis of chronic schizophrenia according to *Diagnostic and Statistical Manual of Mental Disorders* (Fifth edition)[12] criteria were eligible for study participation (**Table 1**)

Table 1. Demographics and Baseline Clinical Characteristics; OLE Safety Population[a]

	AL 441 or 882 mg q4wk (N=478)
Age, mean (SD), years	39.4 (11.6)
Male, n (%)	275 (57.5)
Race, n (%)	
Black or African American	92 (19.2)
White	305 (63.8)
Asian	79 (16.5)
Other	2 (0.4)
Body mass index, mean (SD), kg/m^2	27.0 (5.3)
PANSS total score, mean (SD)[b]	61.5 (14.4)

[a]All patients who received ≥1 dose of AL in OLE study.
[b]Based on patients who received ≥1 dose of AL and had ≥1 postbaseline PANSS assessment in OLE study (n=462). Baseline was defined as the last nonmissing assessment before the first dose of study drug on study day 1 for patients who enrolled de novo or who were assigned to placebo in the lead-in study; baseline was defined as the study day 1 assessment in patients who were assigned to AL in the lead-in study.
AL, aripiprazole lauroxil; OLE, open-label extension; PANSS, Positive and Negative Syndrome Scale.

- Additional eligibility criteria included the following:
 - PANSS total score <70 and Clinical Global Impression–Severity (CGI-S)[13] score ≤3 at screening
 - No hospitalizations for acute exacerbations of schizophrenia ≤3 months before screening

Statistical Analysis

- AL dose groups were combined for this analysis
- Proportions of patients endorsing each response level were summarized for each EQ-5D-5L dimension at 1 month, after all patients had been initiated on a stable dose of AL, and at 1 year
- PSP total scores were summarized descriptively using means (SD) at baseline; mean (SD) total scores and changes from baseline were summarized at 3-month and 1-year time points

RESULTS

Patients

- A total of 478 patients enrolled in the OLE study (**Figure 1**); 462 patients were included in the functional outcomes analysis, and 325 completed 1-year assessments

Figure 1. OLE[a] Patient Flow by Lead-in Group

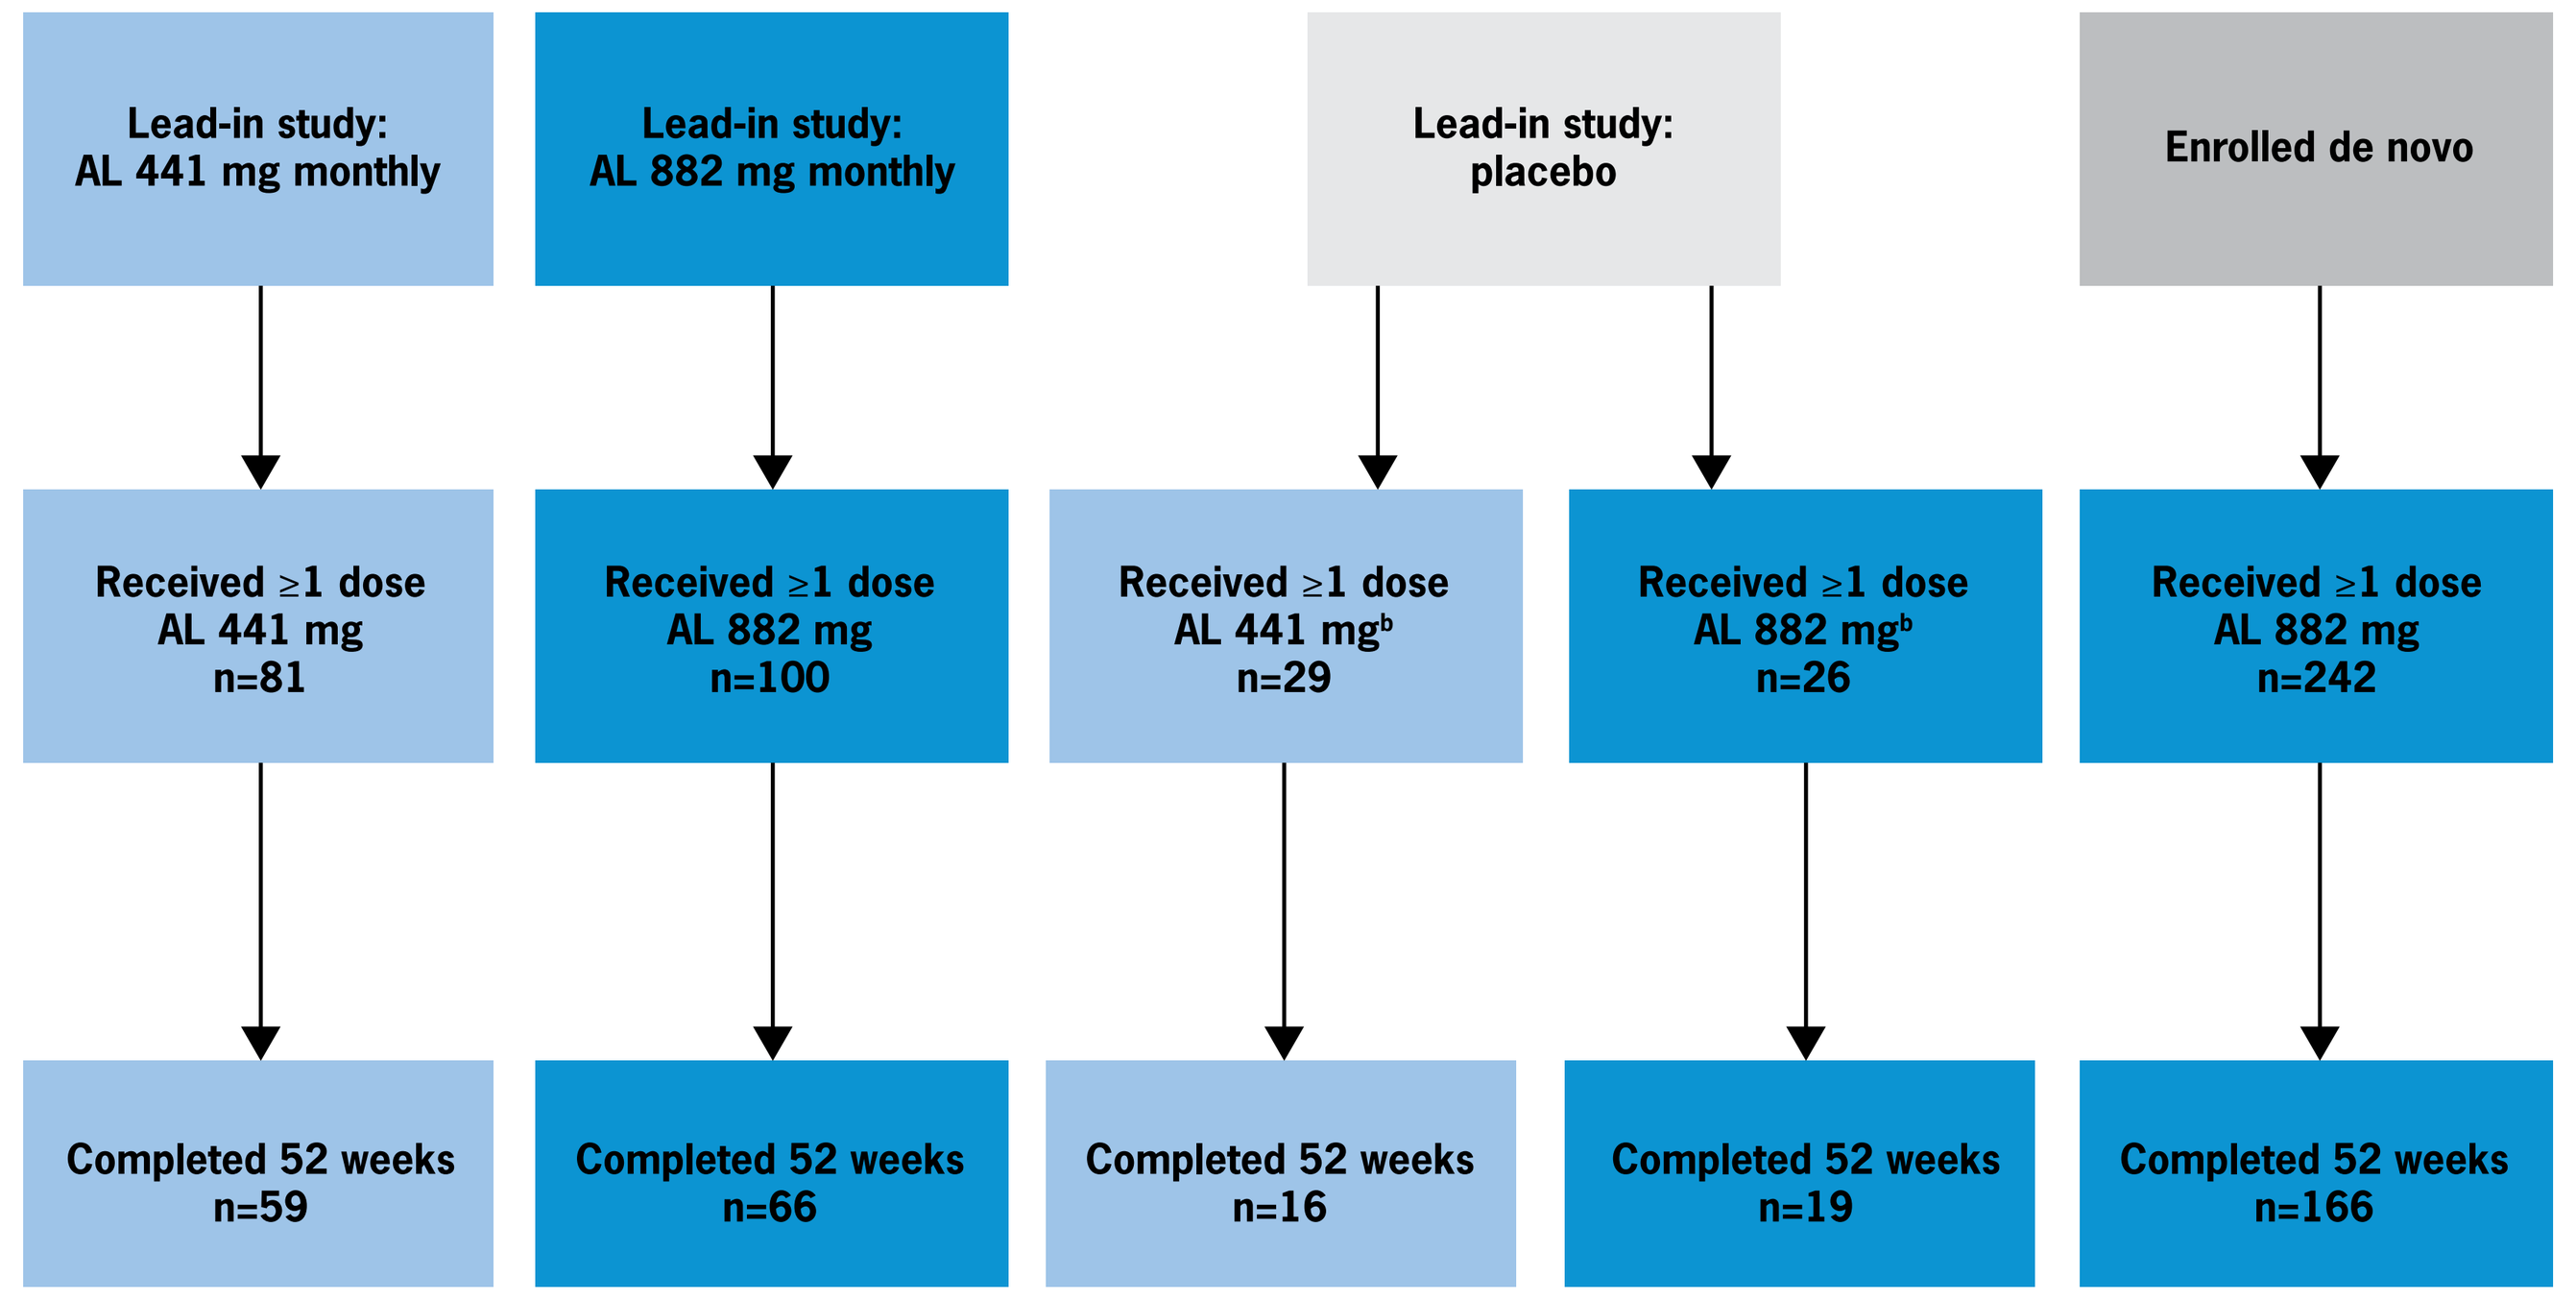
Lead-in study: AL 441 mg monthly
Lead-in study: AL 882 mg monthly
Lead-in study: placebo
Enrolled de novo
Received ≥1 dose AL 441 mg n=81
Received ≥1 dose AL 882 mg n=100
Received ≥1 dose AL 441 mg[b] n=29
Received ≥1 dose AL 882 mg[b] n=26
Received ≥1 dose AL 882 mg n=242
Completed 52 weeks n=59
Completed 52 weeks n=66
Completed 52 weeks n=16
Completed 52 weeks n=19
Completed 52 weeks n=166

[a]Clinicaltrials.gov identifier: NCT01626456.
[b]Patients previously assigned to receive placebo started AL 441 or 882 mg based on their placebo assignment (matched to their 441- or 882-mg placebo injection volume)
AL, aripiprazole lauroxil; OLE, open-label extension.

Health Outcomes

- EQ-5D-5L
 - The proportions of patients reporting “no problems” in functioning ranged from 61% to 85% across the 5 EQ-5D-5L dimensions at 1 month, increasing numerically to 75% to 90% across dimensions at 1 year (**Figure 2**)

Figure 2. Patient-Reported Assessment of Difficulty Functioning in the 5 EQ-5D-5L Domains at 1 Month and 1 Year

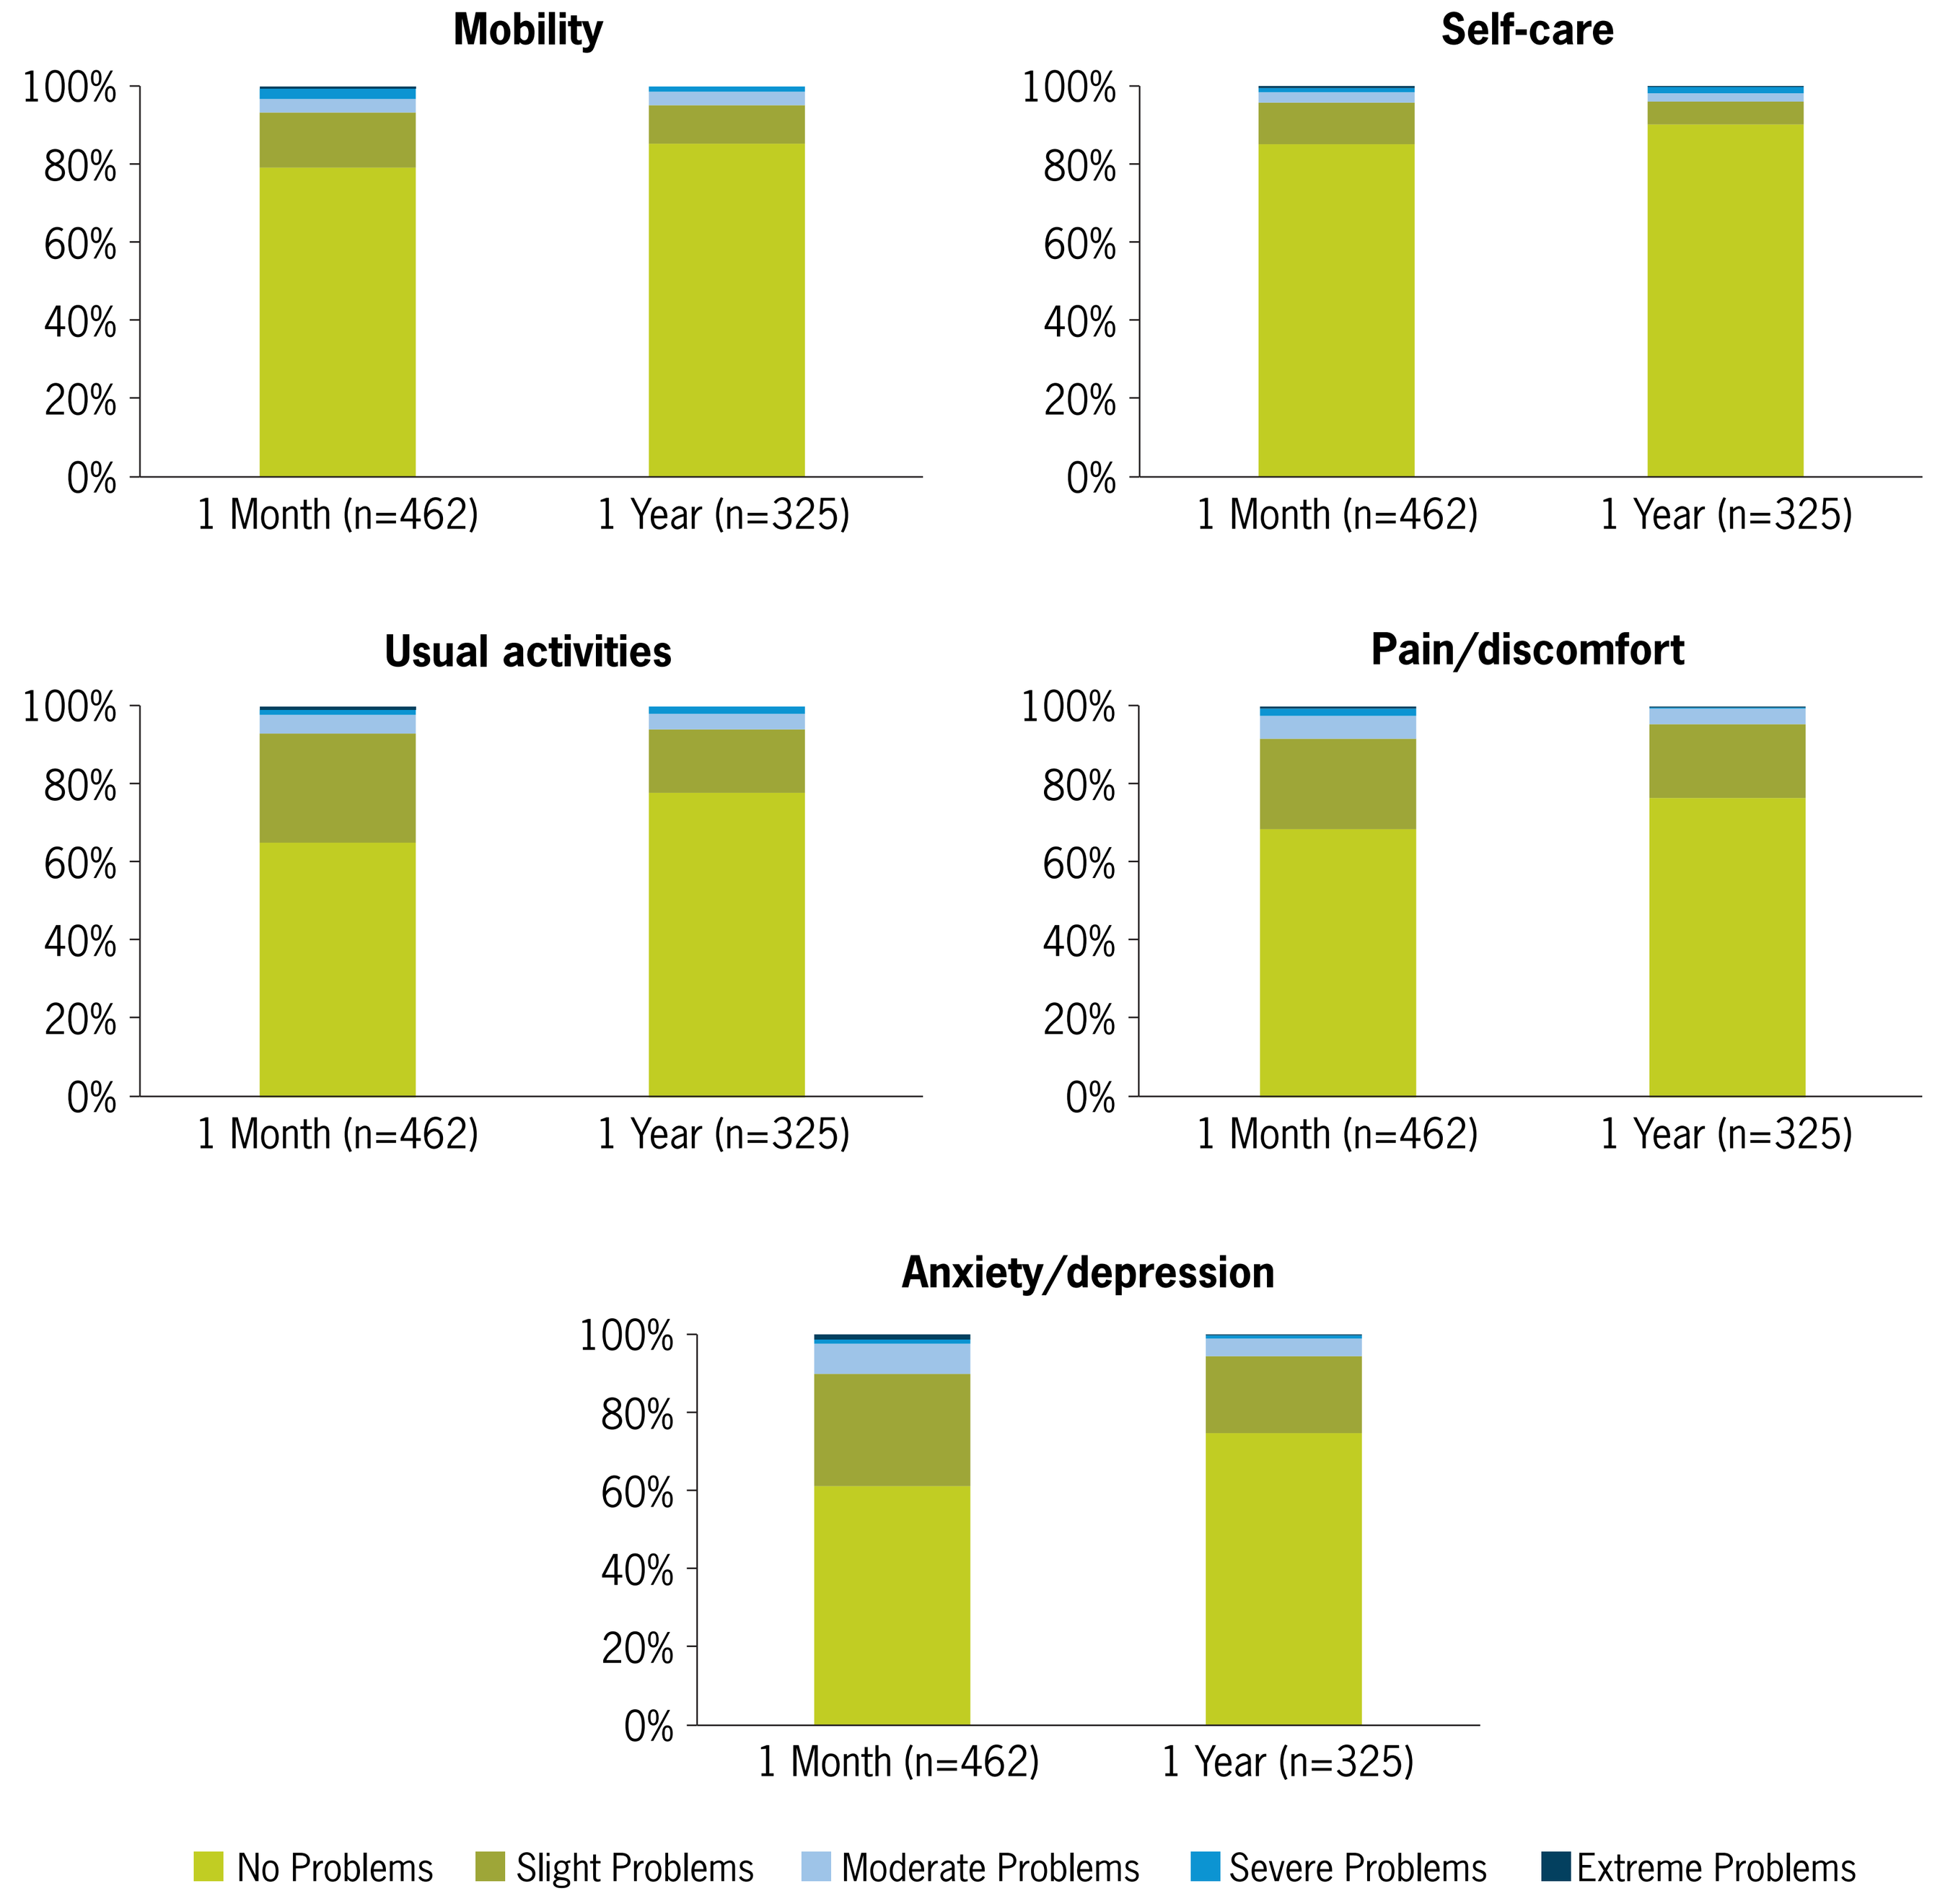
Mobility
Self-care
Usual activities
Pain/discomfort
Anxiety/depression
100%
80%
60%
40%
20%
0%
1 Month (n=462)
1 Year (n=325)
No Problems
Slight Problems
Moderate Problems
Severe Problems
Extreme Problems

- PSP total score
 - Mean PSP total scores on average indicated “manifest but not marked difficulties” (61–70) in functioning at baseline and improved, on average, to “mild difficulties” (71–80) at 1 year (**Table 2**)

Table 2. Clinician-Rated Personal and Social Performance Total Scores[a]

	AL 441 or 882 mg q4wk	
	n	Mean (SD)
Baseline	462	67.7 (12.12)
Month 3	447	69.6 (12.37)
Change from baseline		1.7 (7.17)
1 year	325	74.6 (11.08)
Change from baseline		5.5 (7.89)

[a]Personal and Social Performance total score interpretation: 100–81, good to excellent functioning; 80–71, mild difficulties; 70–61, manifest but not marked difficulties; 60–41, marked difficulties; 40–11, severe difficulties; 10–1, lack of autonomy.
AL, aripiprazole lauroxil.

LIMITATIONS

- This was an open-label AL study with no placebo or active comparator group
- The primary objective of the OLE study was to characterize the safety of AL during long-term treatment of patients with stable schizophrenia; it was not powered to assess changes in functional outcomes
- Because the study population was small and limited to those who met the inclusion and exclusion criteria, these results may not generalize to all patients with schizophrenia and who are treated with antipsychotics
- In addition, early discontinuation by patients with poor response to treatment could have selected for a population with more favorable outcomes

CONCLUSIONS

- The majority (61%-85%) of patients with stable schizophrenia in this OLE study reported "no problems" across EQ-5D-5L health outcome dimensions after 1 month of open-label treatment with the LAI antipsychotic aripiprazole lauroxil
 - The proportion of patients with “no problems” increased from month 1 to 1 year during extended aripiprazole lauroxil treatment
- Numerical improvement in functioning over 1 year of extended treatment with aripiprazole lauroxil also was observed on the clinician-reported PSP scale
- These results suggest that long-term, consistent treatment with aripiprazole lauroxil may be associated with improvements in self-reported health-related quality of life, from the patient and clinician perspectives

REFERENCES

1. Harvey PD, et al. *Schizophr Res*. 2012;140:1-8. **2.** Lambert M, et al. *Dialogues Clin Neurosci*. 2010;12:393-407. **3.** Karadayi G, et al. *Compr Psychiatry*. 2011;52:701-7. **4.** Lambert M, et al. *J Clin Psychiatry*. 2006;67:1690-7. **5.** Lahera G, et al. *BMC Psychiatry*. 2018;18:176. **6.** Kay SR, et al. *Schizophr Bull*. 1987;13:261-76. **7.** Meltzer HY, et al. *J Clin Psychiatry*. 2015;76:1085-90. **8.** McEvoy JP, et al. *J Clin Psychiatry*. 2017;78:1103-9. **9.** Nasrallah HA, et al. *CNS Spectr*. 2019;24:395-403. **10.** Herdman M, et al. *Qual Life Res*. 2011;20:1727-36. **11.** Morosini PL, et al. *Acta Psychiatr Scand*. 2000;101:323-9. **12.** American Psychiatric Association. Diagnostic and Statistical Manual of Mental Disorders, Fifth Edition. Washington, DC: American Psychiatric Publishing; 2013. **13.** Guy W. ECDEU Assessment Manual for Psychopharmacology. Revised. Rockville, MD: US Department of Health, Education and Welfare; 1976.

AUTHOR DISCLOSURES

SG, SD, MW, and BDR are employees of Alkermes, Inc., and may be shareholders.

ACKNOWLEDGMENTS

Medical writing and editorial support were provided by Peloton Advantage, LLC, an OPEN Health company, and funded by Alkermes, Inc.

Copies of this poster obtained through this QR (Quick Response) code are for personal use only and may not be reproduced without permission of Alkermes, Inc. For permission, contact USMedInfo@Alkermes.com.

Supported by funding from Alkermes, Inc.