Diagnosis and Treatment Patterns of GERD in Non-Gastrointestinal Departments In China: a Subgroup Analysis From a Retrospective Real-World Study (GRAND China)

HSD78

Xiaoyun Yu[1], Li Xie[2], Qi Song[2], Xiaohua Hou[1]*

[1]Division of Gastroenterology, Union Hospital affiliated with Tongji Medical College of Huazhong University of Science and Technology, Wuhan, China; [2]Medical Affairs, Takeda Pharmaceutical Company, Beijing, China

*Corresponding author

INTRODUCTION

- Patients with gastroesophageal reflux disease (GERD), a chronic condition characterized by abnormal reflux of gastric contents into the esophagus, exhibit disruptive symptoms and complications, including esophageal and extraesophageal manifestations[1,2]
- Reportedly, the pooled prevalence of GERD in China based on 10 studies is 4.16%,[3] with an increasing trend[4]
- The diagnosis of GERD is challenging in the absence of the typical symptoms, heartburn and regurgitation[2]
- Limited evidence exists to understand the challenges when presented with atypical or extraesophageal symptoms and a potential GERD diagnosis in non-gastrointestinal (GI) departments

OBJECTIVE

- To describe the diagnosis, symptom subtypes, and treatment patterns of GERD detected in non-GI departments in China

METHODS

Study design and data collection

- This was a subgroup analysis of the retrospective, real-world GRAND China study using the pooled data of patients with a GERD diagnosis
- Data were extracted from electronic medical records (January 2015 to December 2017) of 11 hospitals across 10 provinces in a standardized format and analyzed after de-identification
- The study was approved by the Institutional Ethics Committee of Tongji Medical College of Huazhong University of Science and Technology

Eligibility criteria

- Patients of all ages identified using a primary diagnosis (i.e., among the top two diagnoses) of GERD during their outpatient visit per the International Classification of Diseases, Tenth Revision codes, including K20xx05, K20xx06, K21.001, K21.901, K21.902, K21.903, K21.904, K22.103, and K22.105, or using the medical terms "esophageal reflux" and "esophagitis"

Outcomes

- Symptoms assessed were
 - Typical (heartburn, regurgitation)
 - Atypical (chest pain, dysphagia, epigastric pain, belching, bloating, nausea, vomiting)
 - Extraesophageal (globus sensation, cough, asthma, laryngitis, dental erosion)
- Non-GI departments included departments of otolaryngology (ENT), respiratory, cardiovascular (CV), and others such as general medicine, general surgery, and traditional Chinese medicine
- Treatment pattern assessments included the following:
 - Mode of treatment: prescribed medicines used alone or in combination
 - Type of medication: proton pump inhibitors (PPIs), prokinetic agents, anti-acid agents/mucosal protectants, or histamine 2 receptor (H_2) antagonists
 - Dose: standard dose, lower-than-standard dose, or higher-than-standard dose
 - Frequency: once daily, twice daily, thrice daily

Statistical analysis

- Data processing and statistical analyses were performed using R-3.6.2 software (R Foundation for Statistical Computing, Vienna, Austria)
- Descriptive analyses are reported as frequency and percentage for categorical variables

RESULTS

Patient disposition

- Of the 71,904 patients diagnosed with GERD, 34.4% were diagnosed in non-GI departments (14.0%, ENT; 0.9%, respiratory; 0.2%, CV; and 19.3%, others; **Figure 1**)
- The mean (standard deviation) age was 49.3 (14.6) years
- The study population included similar proportions of men (n=34,601; 48.1%) and women (n=37,279; 51.8%)

Figure 1. Patient disposition

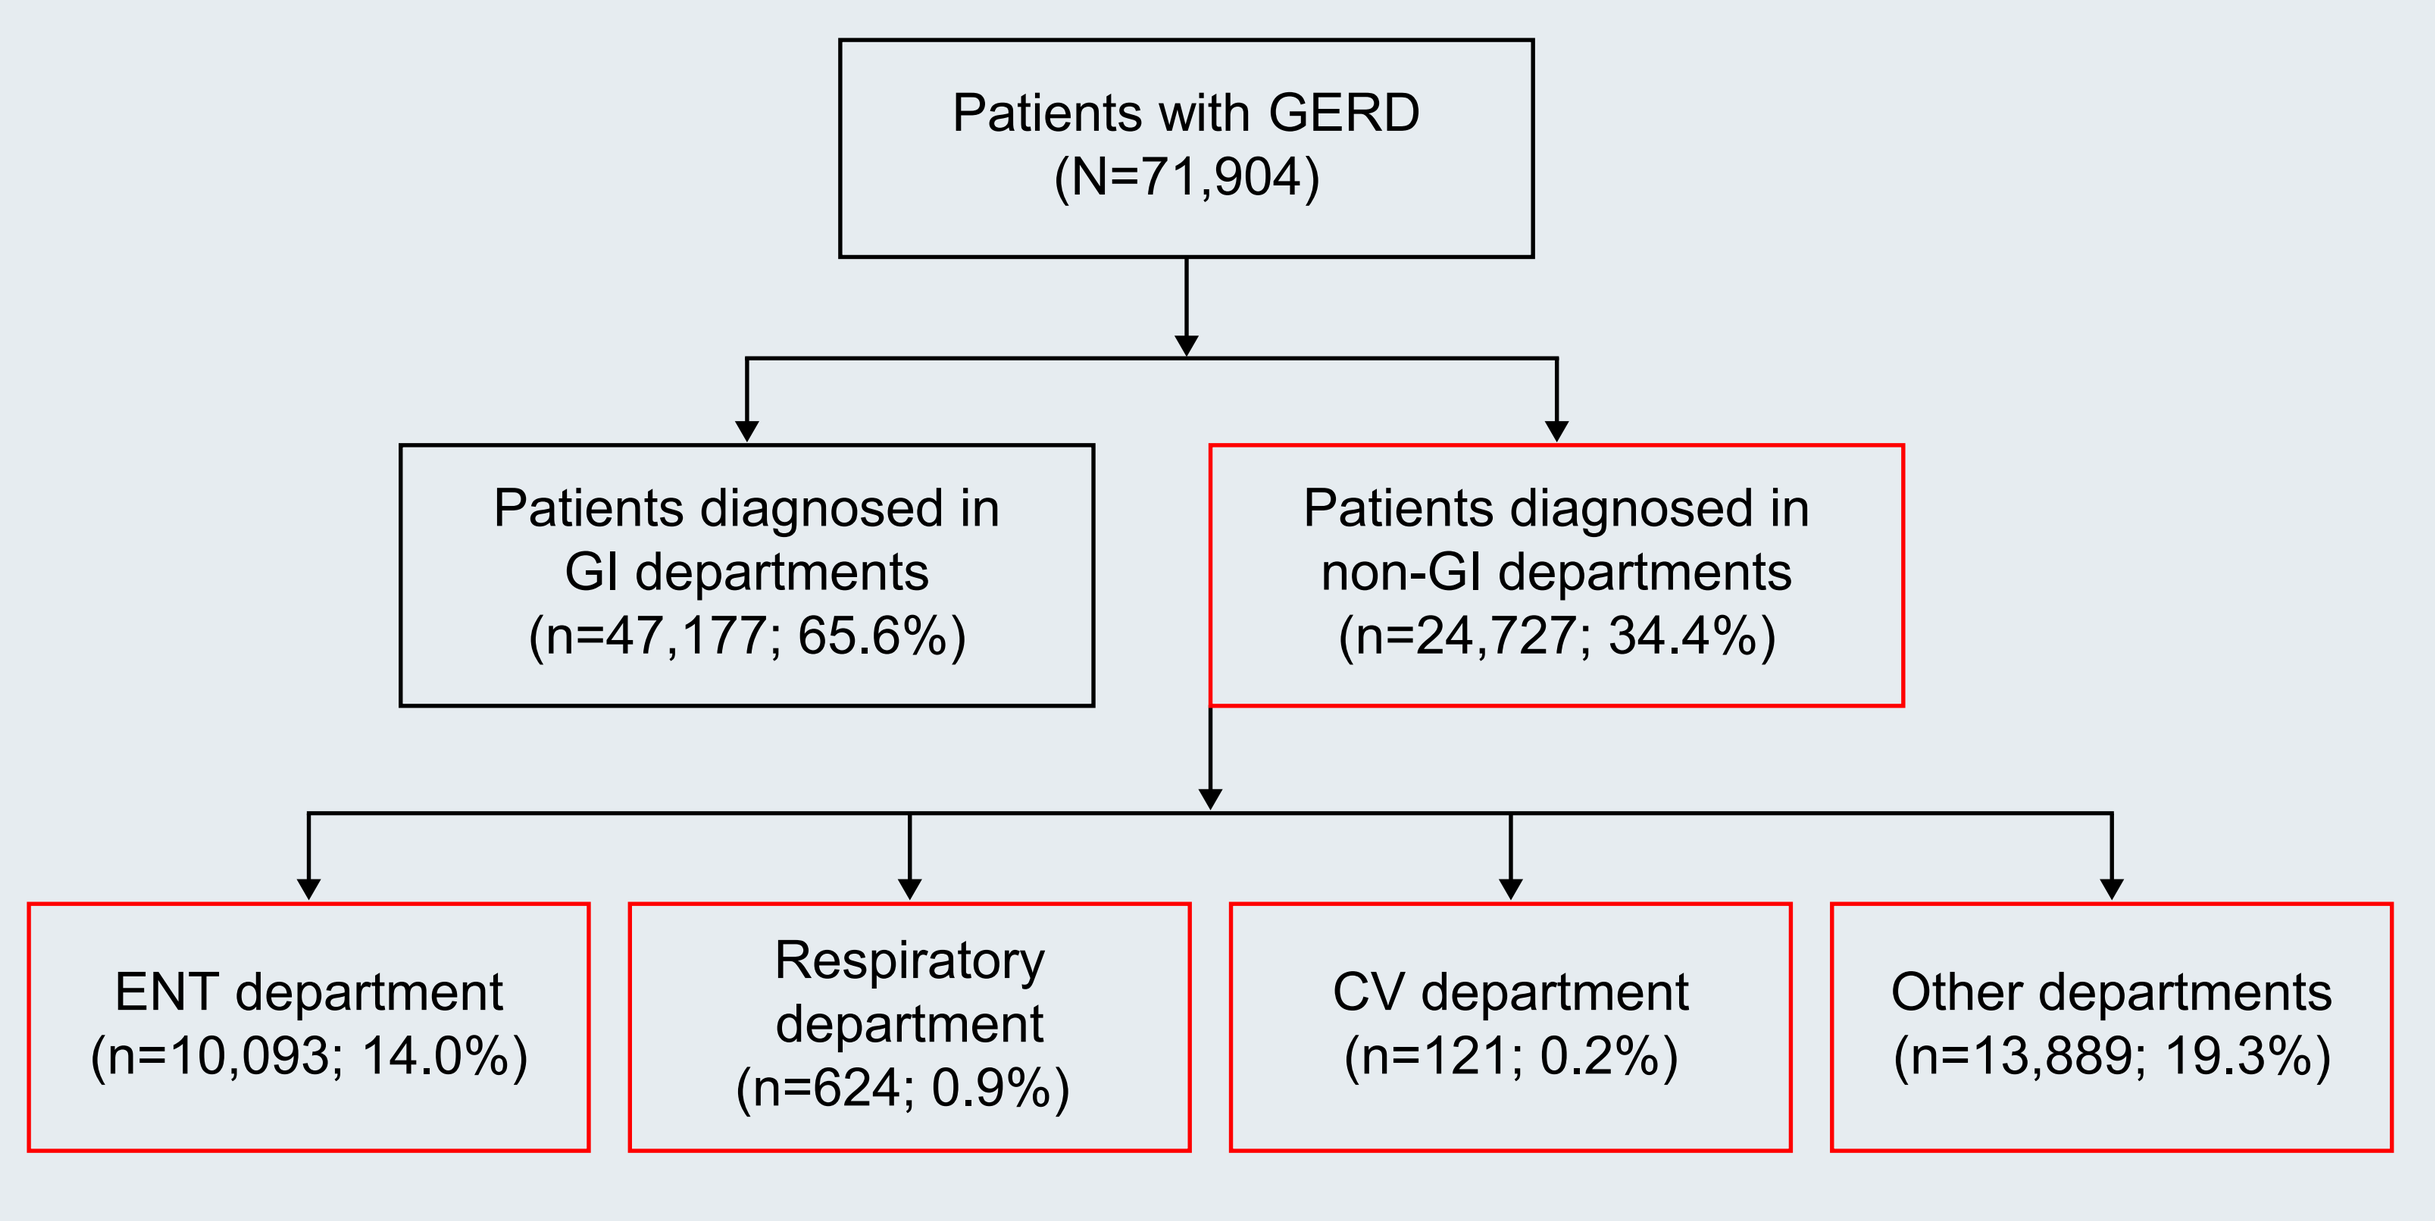
Patients with GERD
(N=71,904)
Patients diagnosed in GI departments (n=47,177; 65.6%)
Patients diagnosed in non-GI departments (n=24,727; 34.4%)
ENT department (n=10,093; 14.0%)
Respiratory department (n=624; 0.9%)
CV department (n=121; 0.2%)
Other departments (n=13,889; 19.3%)

Other departments included general medicine, general surgery, and traditional Chinese medicine. CV, cardiovascular; ENT, otolaryngology; GERD, gastroesophageal reflux disease; GI, gastrointestinal.

Diagnosis trends by type of department

- Between 2015 and 2017, the number of diagnoses in the ENT department showed a steady increase (2015, 10.5%; 2016, 13.8%; and 2017, 17.6%), and those in the respiratory (1.0%, 0.9%, and 0.7%, respectively) and CV (0.1%, 0.2%, and 0.2%, respectively) departments remained stable, whereas those in other departments (22.9%, 19.0%, and 16.3%, respectively) showed decreasing trends (**Figure 2**)
- Patients reported typical (overall, 35.3%; ENT, 17.0%; respiratory, 12.3%; CV, 23.1%; others, 40.8%), atypical (overall, 40.9%; ENT, 13.2%; respiratory, 26.5%; CV, 84.6%; others, 44.5%), and extraesophageal (overall, 35.6%; ENT, 71.7%; respiratory, 68.8%; CV, 0.0%; others, 27.7%) symptoms (**Figure 3**)

Figure 2. Diagnosis patterns by type of department during the study period (2015–2017)

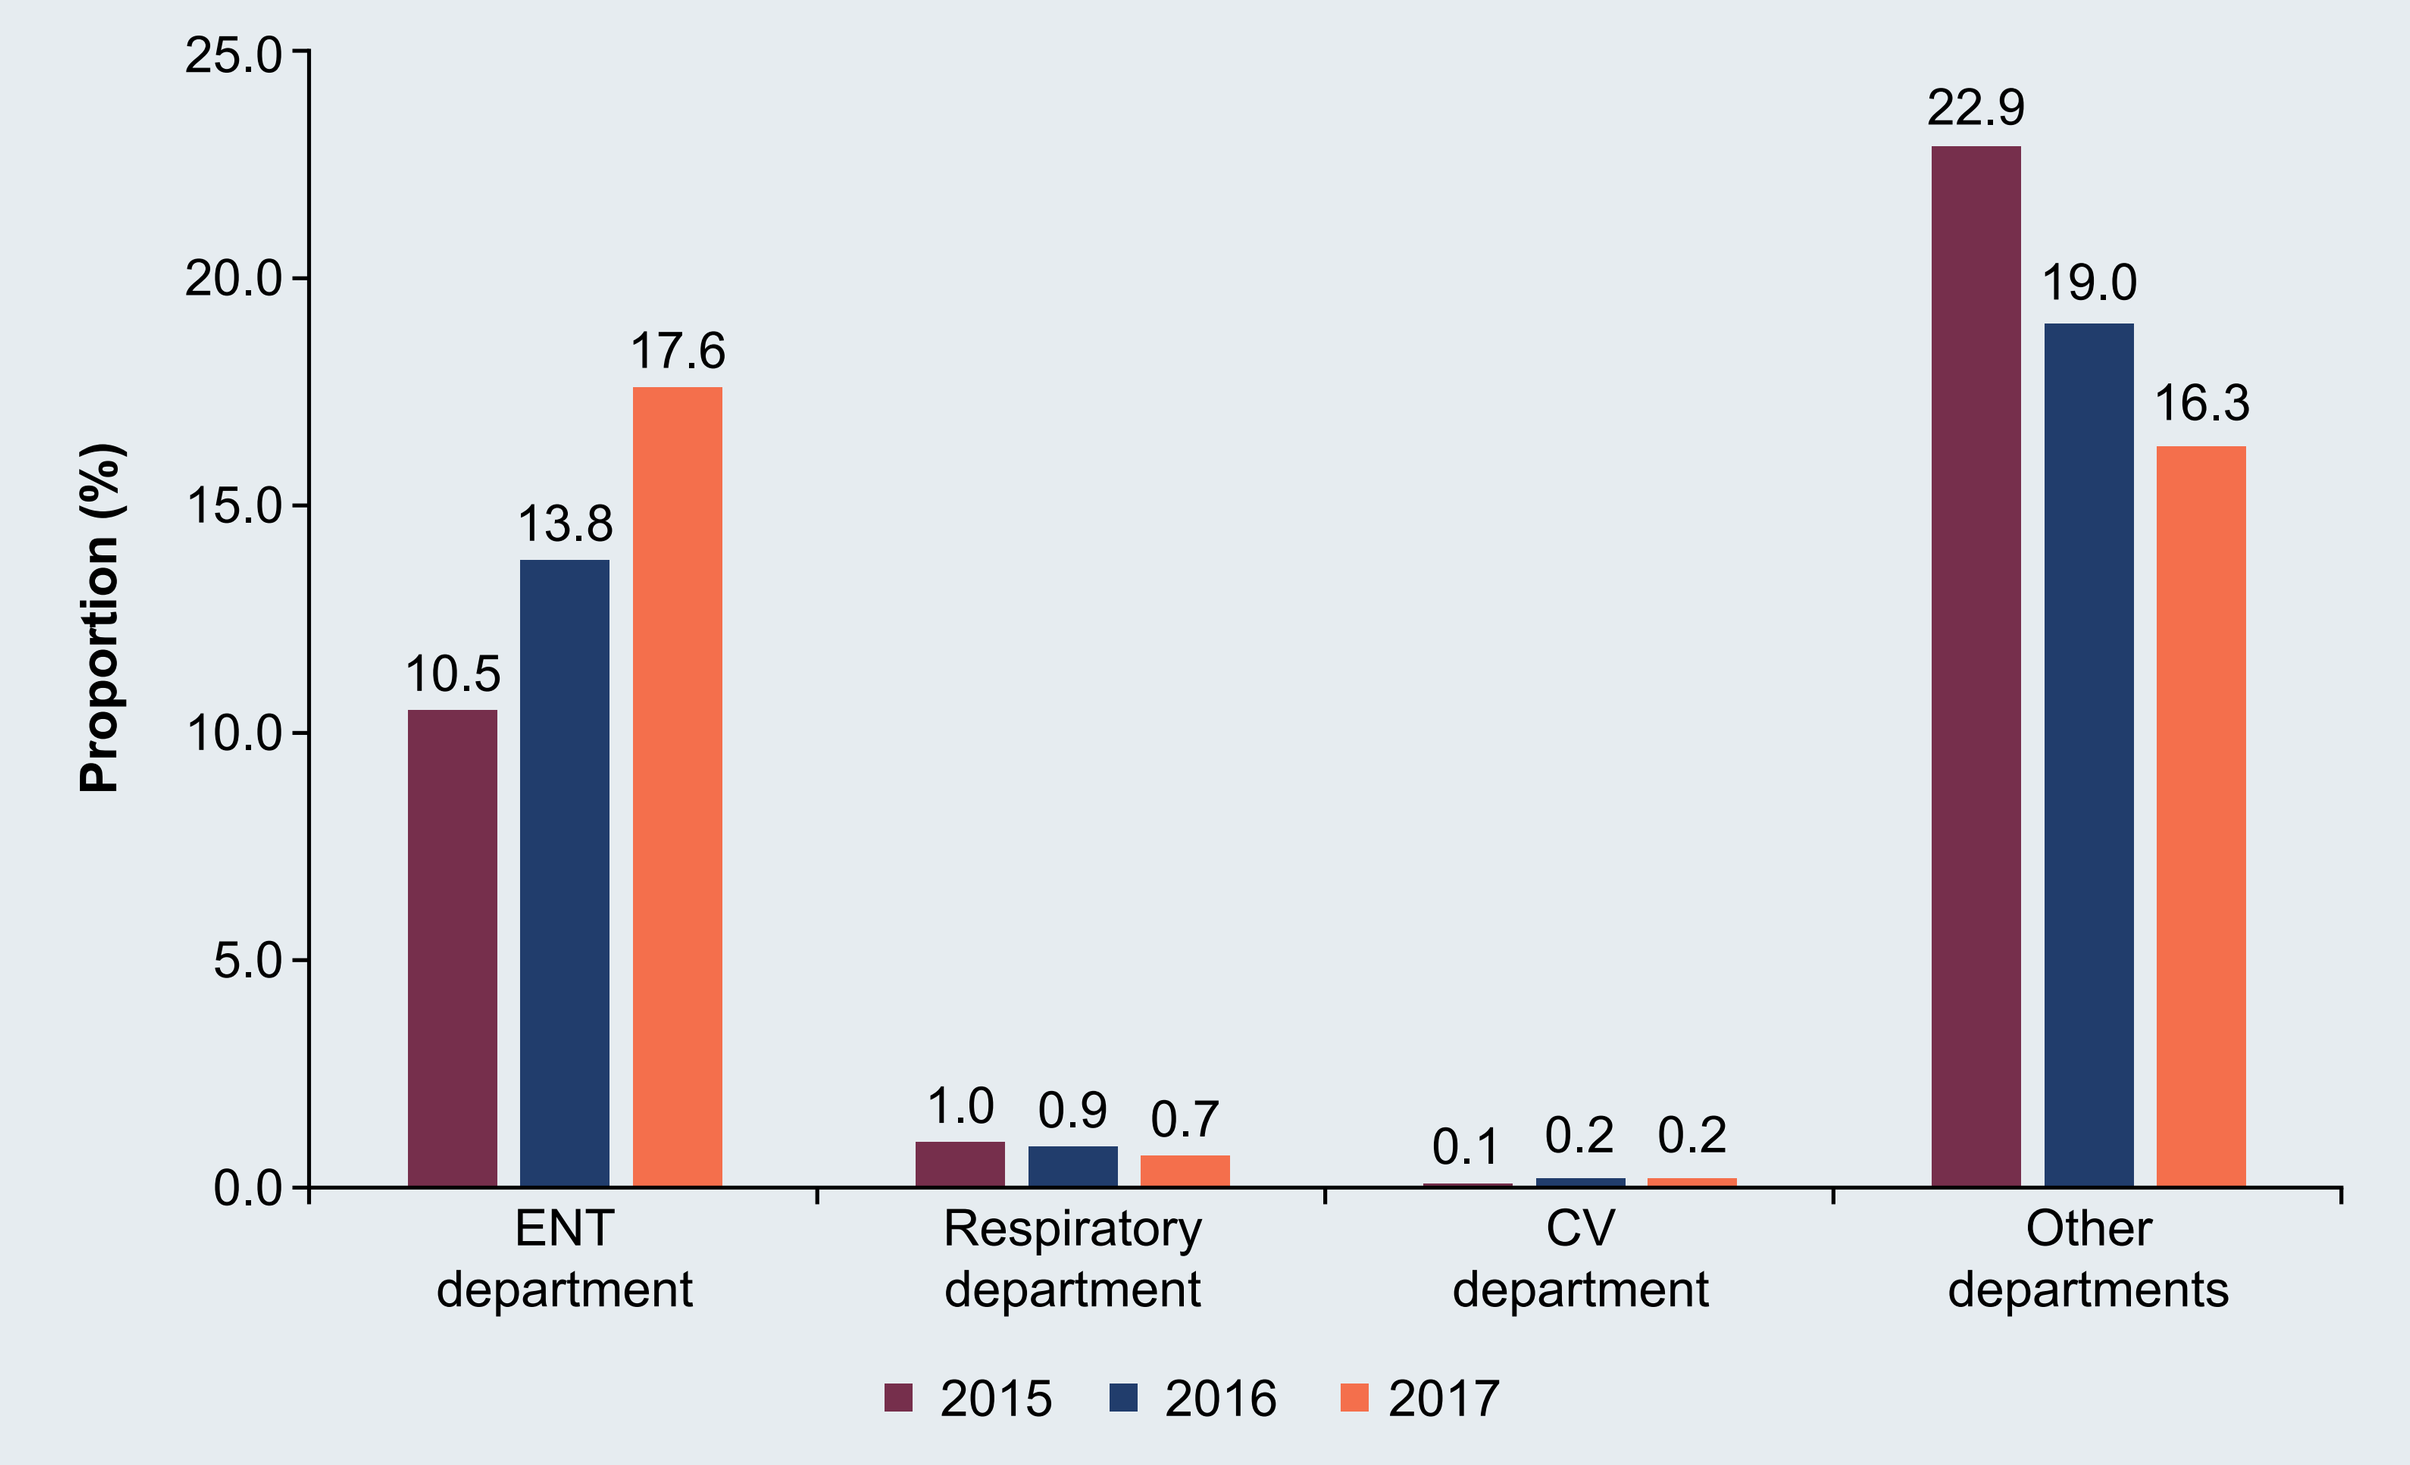
Proportion (%)
25.0
20.0
15.0
10.0
5.0
0.0
10.5
13.8
17.6
1.0
0.9
0.7
0.1
0.2
0.2
22.9
19.0
16.3
ENT department
Respiratory department
CV department
Other departments
2015
2016
2017

Other departments included general medicine, general surgery, and traditional Chinese medicine. CV, cardiovascular; ENT, otolaryngology.

Figure 3. Distribution of symptom subtypes by type of department

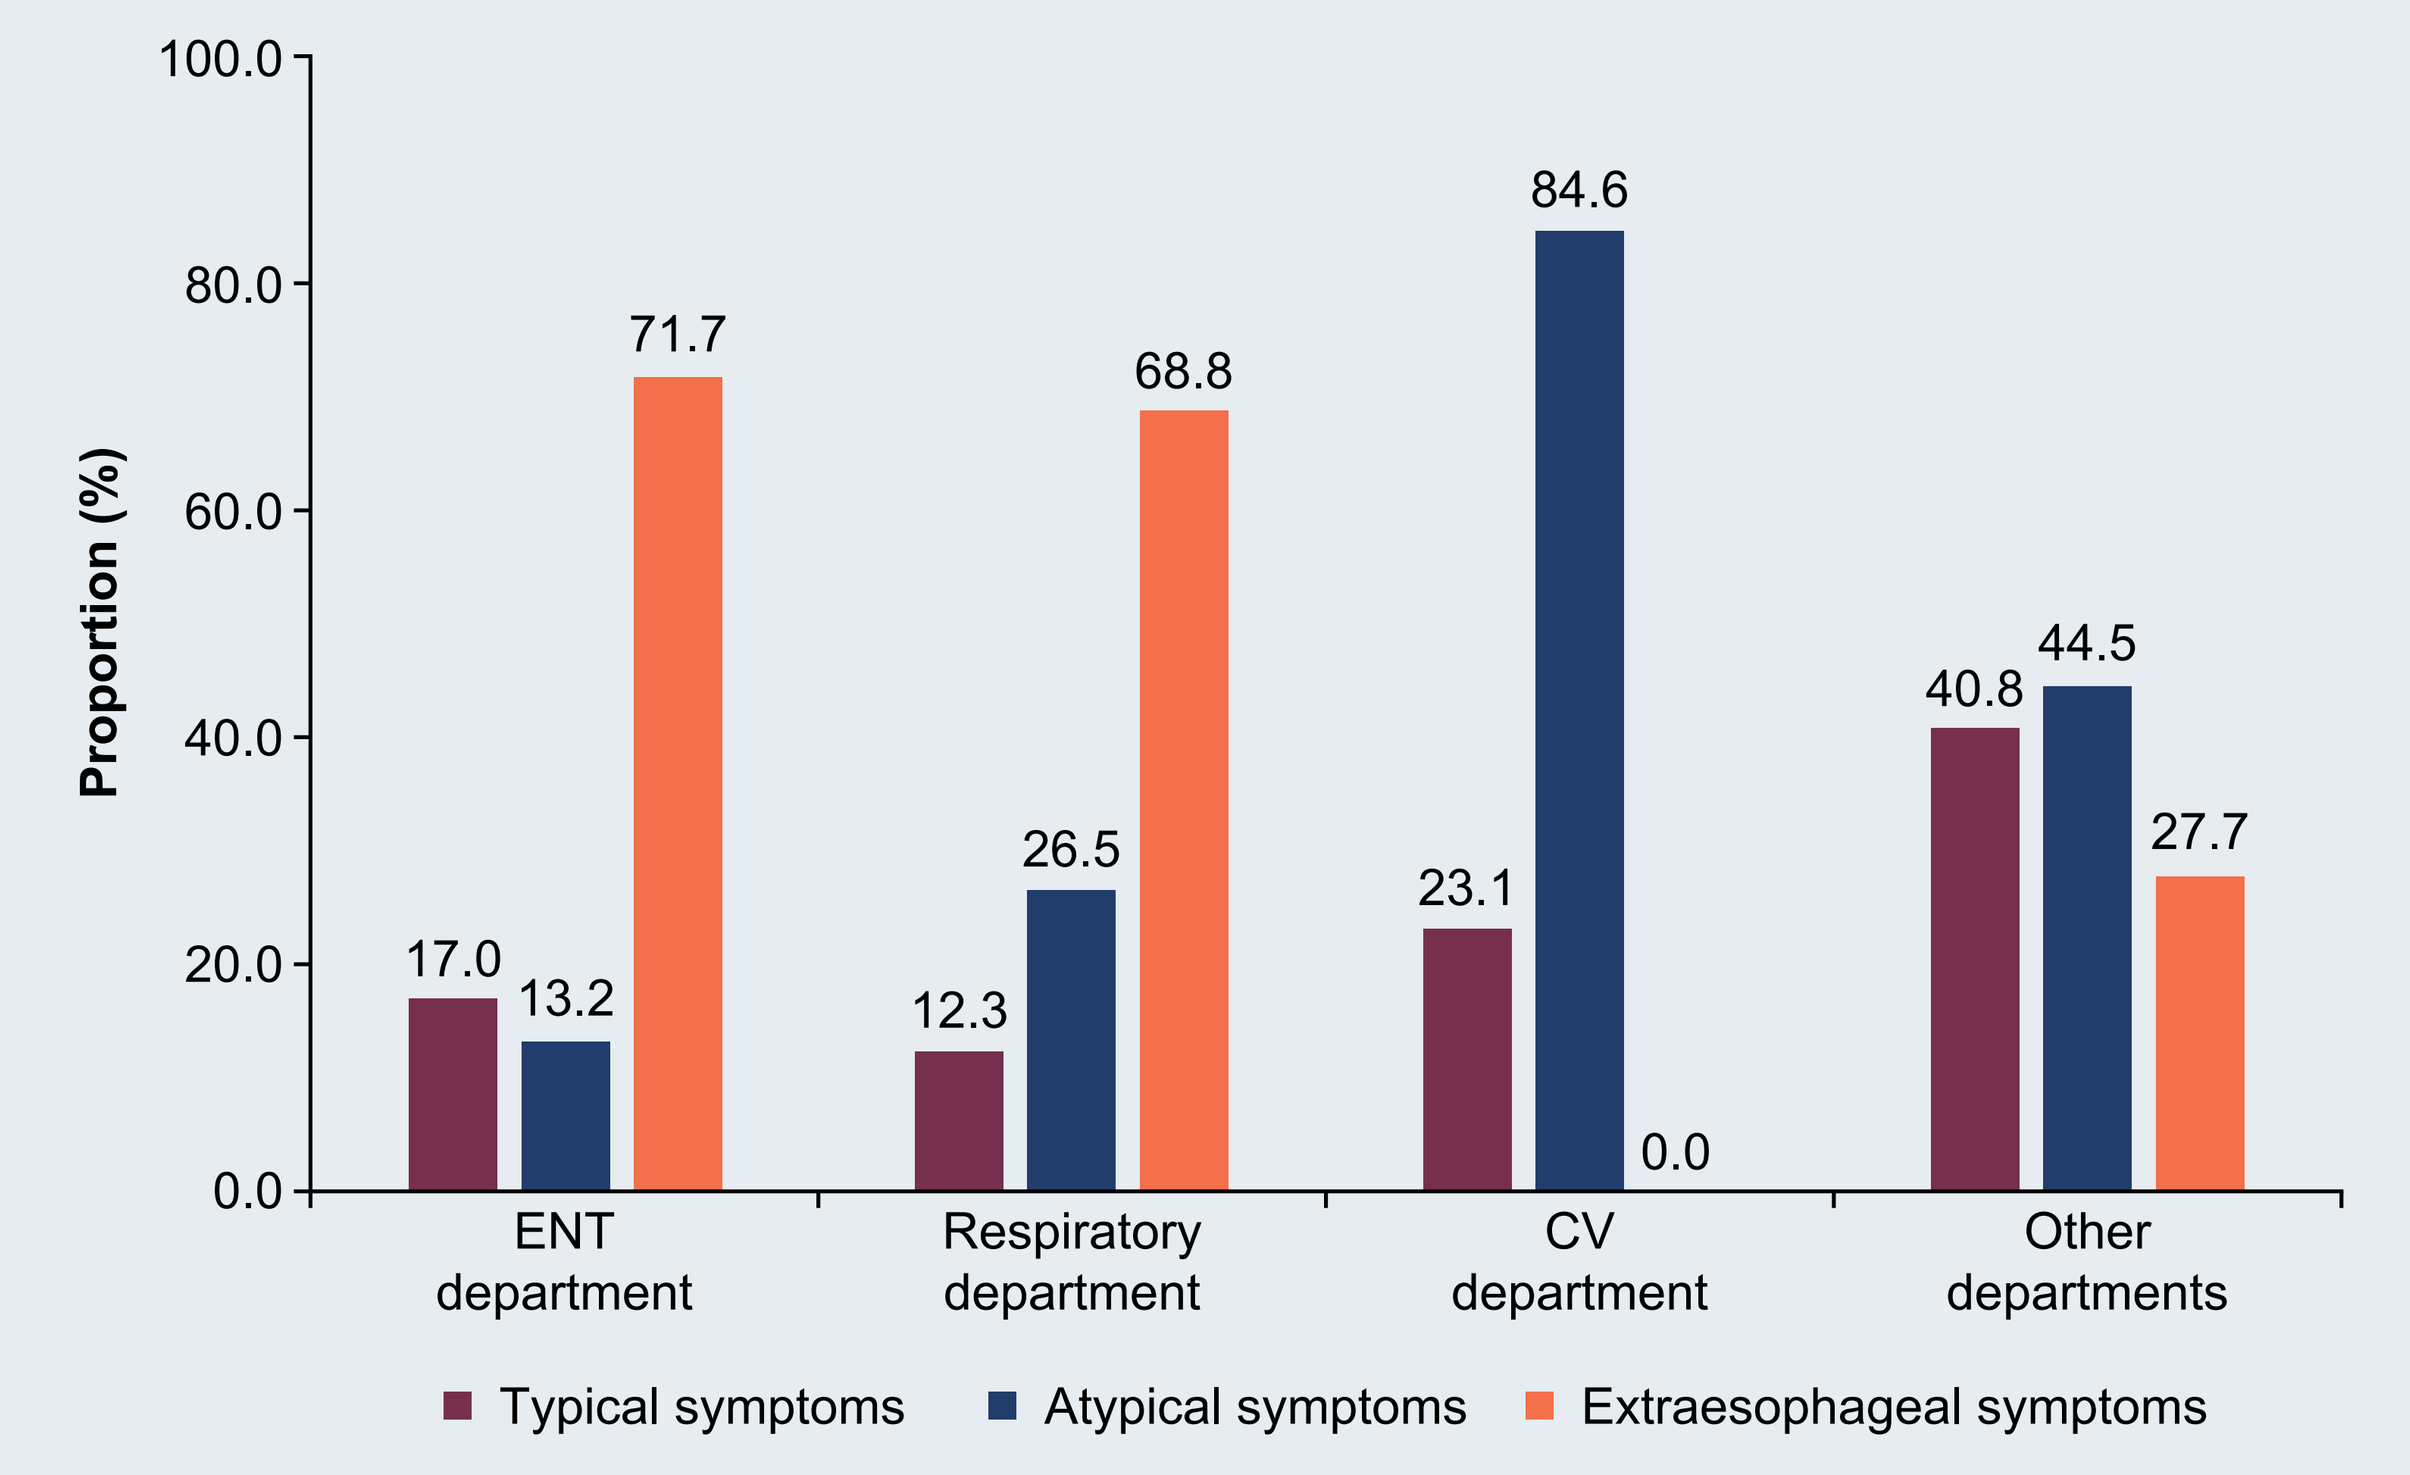
Proportion (%)
100.0
80.0
60.0
40.0
20.0
0.0
17.0
13.2
71.7
12.3
26.5
68.8
23.1
84.6
0.0
40.8
44.5
27.7
ENT department
Respiratory department
CV department
Other departments
Typical symptoms
Atypical symptoms
Extraesophageal symptoms

Other departments included general medicine, general surgery, and traditional Chinese medicine. CV, cardiovascular; ENT, otolaryngology.

Treatment patterns

- Therapies were prescribed as combination therapy (66.1%) or monotherapy (33.9%)
- PPIs were most prescribed (94.0%), followed by prokinetic agents (57.5%), anti-acid agents/mucosal protectants (24.4%), and H_2 antagonists (2.4%; **Figure 4**)
 - PPIs were mostly prescribed in combination with prokinetic agents (32.2%) or prokinetic agents + anti-acid agents/mucosal protectants (8.2%; **Table 1**)
- Most prescriptions were for higher-than-standard doses (47.9%); 31.6% were for the twice-daily dosing frequency (**Figure 5**)

Figure 4. Treatment patterns by therapies

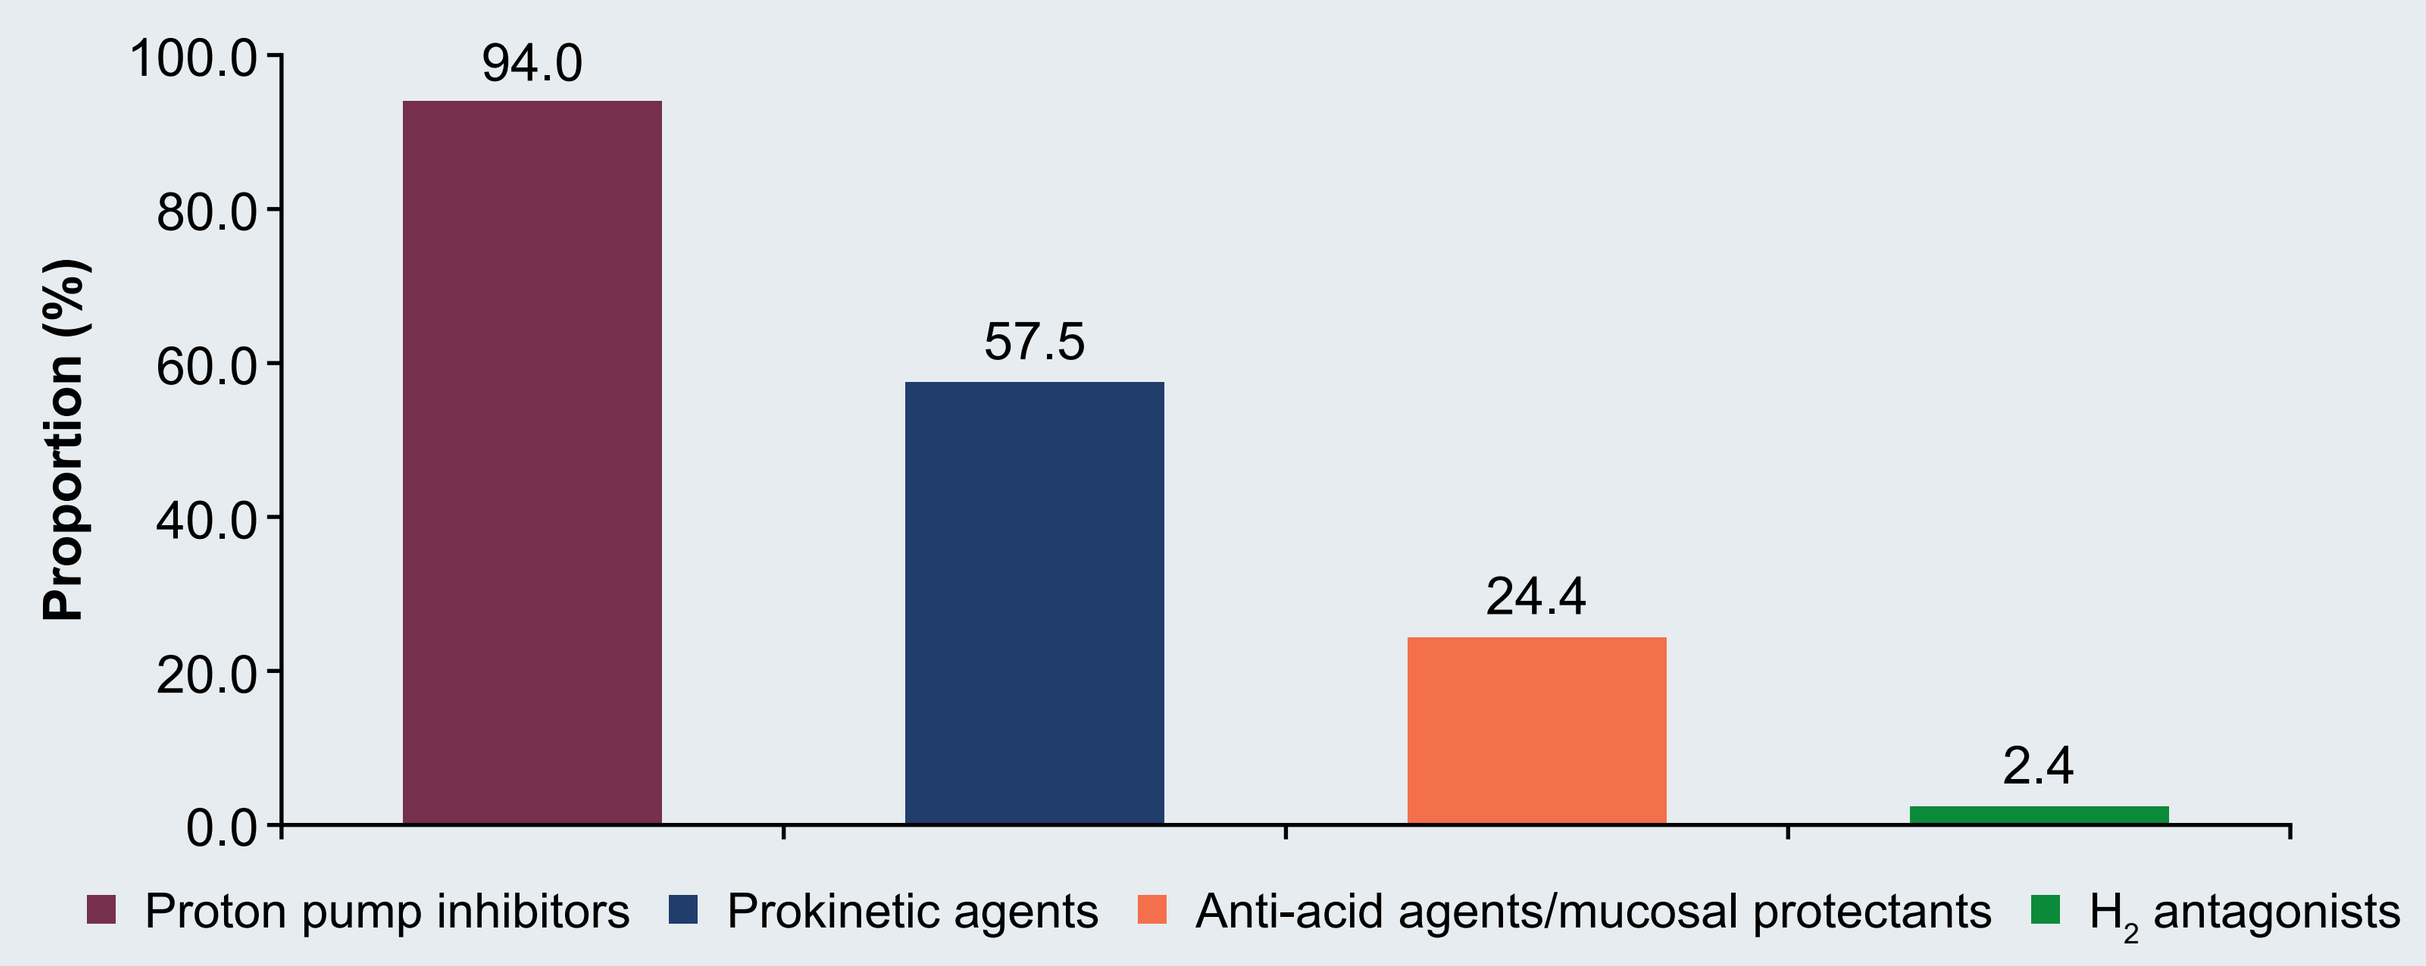
Proportion (%)
100.0
80.0
60.0
40.0
20.0
0.0
94.0
57.5
24.4
2.4
Proton pump inhibitors
Prokinetic agents
Anti-acid agents/mucosal protectants
H2 antagonists

H_2 antagonist, histamine 2 receptor antagonist.

Table 1. Combination therapies prescribed for patients with GERD

Therapy combination	Proportion, n (%)
Combination	
PPIs + prokinetic agents	10,279 (32.2)
Anti-acid agents/mucosal protectants + PPIs	2497 (7.8)
Anti-acid agents/mucosal protectants + prokinetic agents	142 (0.4)
PPIs + H_2 antagonists	62 (0.2)
Prokinetic agents + H_2 antagonists	84 (0.3)
Anti-acid agents/mucosal protectants + H_2 antagonists	35 (0.1)
Triple	
Anti-acid agents/mucosal protectants + PPIs + prokinetic agents	2624 (8.2)
PPIs + prokinetic agents + H_2 antagonists	182 (0.6)
Anti-acid agents/mucosal protectants + PPIs + H_2 antagonists	25 (0.1)
Anti-acid agents/mucosal protectants + prokinetic agents + H_2 antagonists	19 (0.1)
Quadruple	
Anti-acid agents/mucosal protectants + PPIs + prokinetic agents + H_2 antagonists	55 (0.2)

GERD, gastroesophageal reflux disease; H_2 antagonist, histamine 2 receptor antagonist; PPI, proton pump inhibitor.

Figure 5. Dose and frequency of therapies

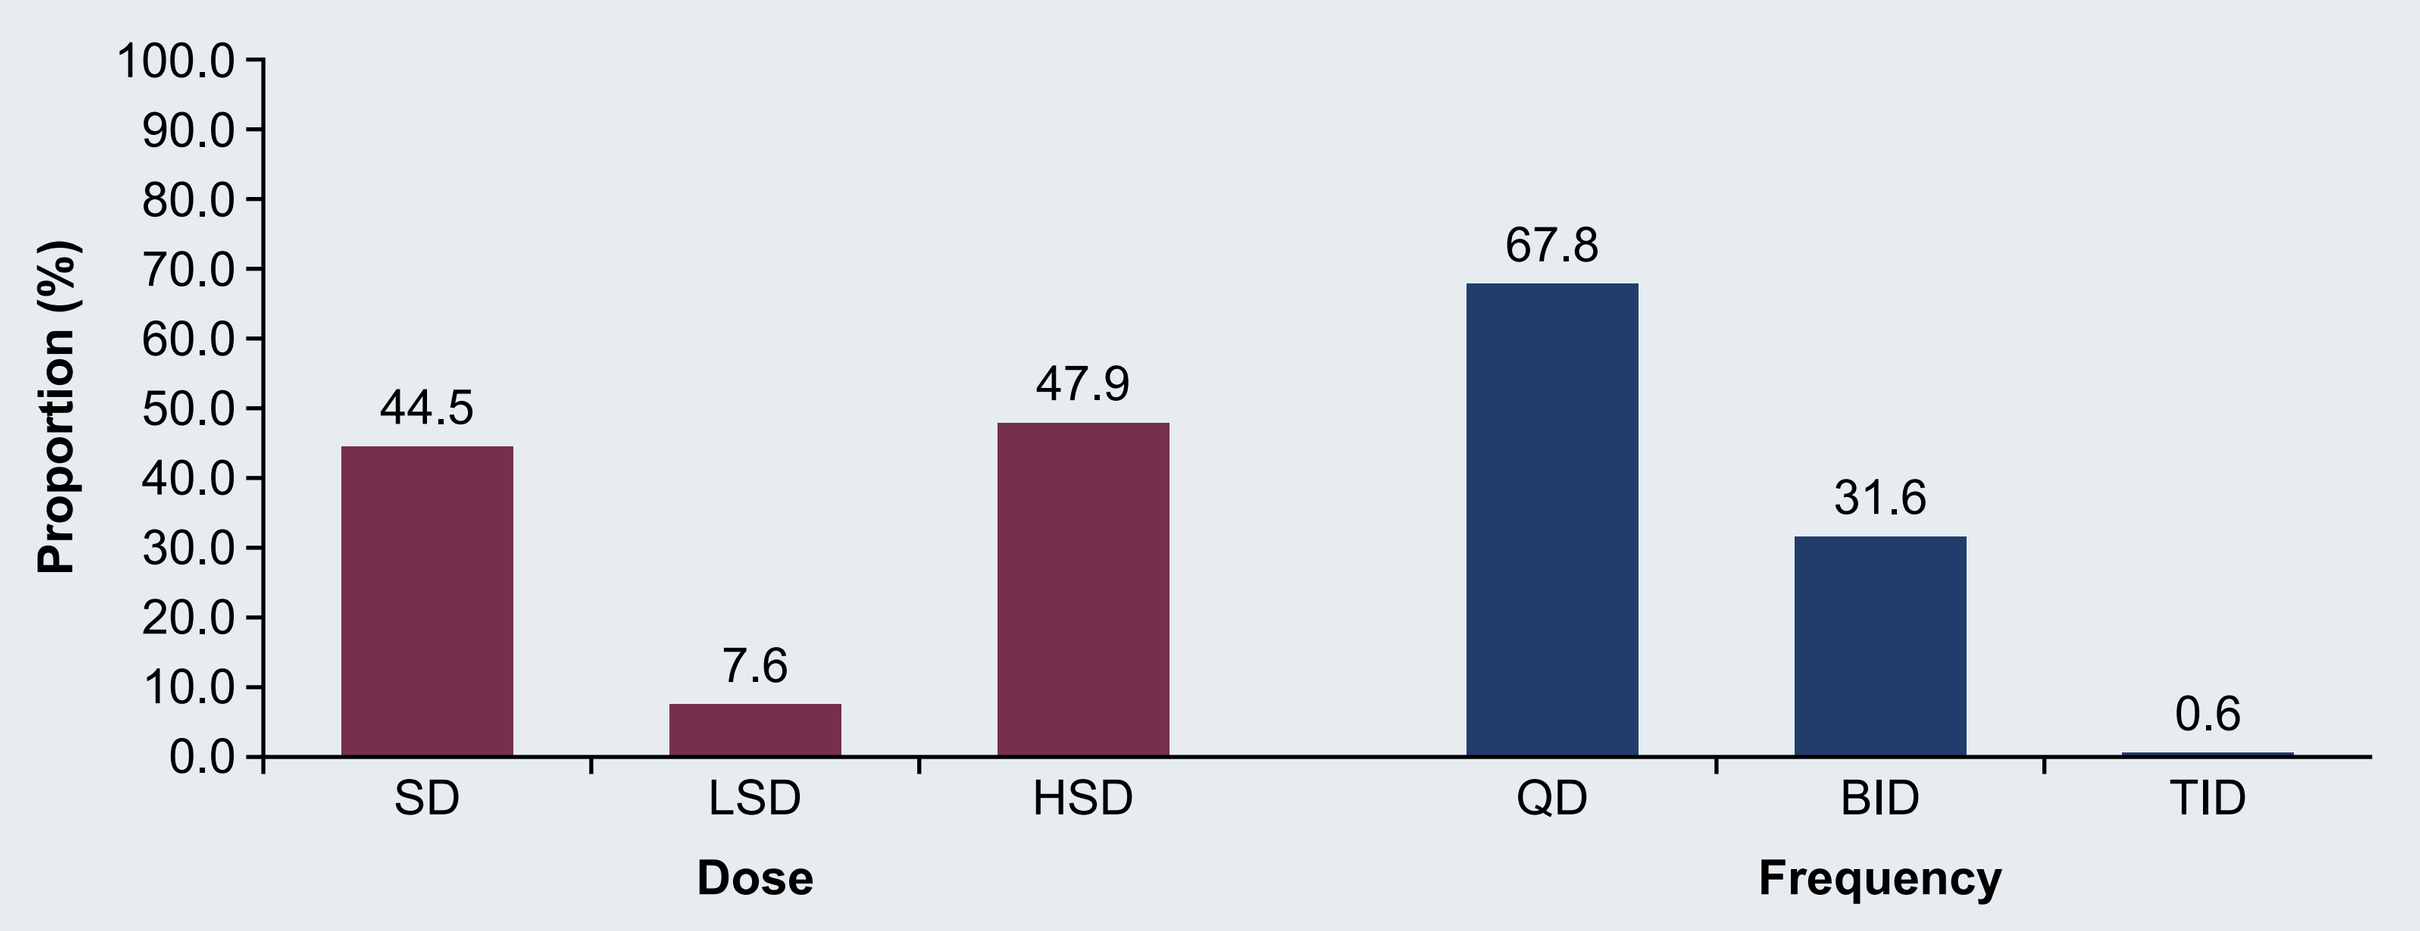
Proportion (%)
100.0
90.0
80.0
70.0
60.0
50.0
40.0
30.0
20.0
10.0
0.0
44.5
7.6
47.9
67.8
31.6
0.6
SD
LSD
HSD
QD
BID
TID
Dose
Frequency

BID, twice daily; HSD, higher-than-standard dose; LSD, lower-than-standard dose; QD, once daily; SD, standard dose; TID, thrice daily.

LIMITATIONS

- Retrospective study design and heterogeneity in data reporting

CONCLUSIONS

- Patients with GERD are diagnosed in non-GI departments based on the presentation of atypical and extraesophageal symptoms
- There is an increasing trend in the diagnosis of GERD in ENT departments
- The results indicate unmet needs in the treatment of GERD in non-GI departments based on treatment intensification, as shown by the prescription of combination therapy, higher-than-standard dose, and twice-daily frequency

REFERENCES

1. Vakil N, et al. *Am J Gastroenterol* 2006;101:1900–20.
2. Clarrett DM, Hachem C. *Mo Med* 2018;115:214–8.
3. Nirwan JS, et al. *Sci Rep* 2020;10:5814.
4. Tan VP, et al. *J Clin Gastroenterol* 2016;50:e1–7.

DISCLOSURES

- The study was funded by Takeda Pharmaceutical Company, China
- XY and XH have no conflicts of interest
- LX is an employee of Takeda Pharmaceutical Company, China, and holds Takeda stock options
- QS is an employee of Takeda Pharmaceutical Company, China

ACKNOWLEDGMENTS

- The authors thank the site investigators and their research teams for participating in and contributing to the GRAND China study
- Medical writing support for the development of this poster was provided by Shruti Muralidharan, PhD, of Cactus Life Sciences (part of Cactus Communications) in accordance with Good Publication Practice Guidelines (https://www.ismpp.org/gpp3) and funded by Takeda Pharmaceutical Company Limited (China)

This poster is intended for healthcare providers only. Presented at ISPOR 2022 (May 15–18, 2022)