Patient Preferences for Attributes of Advanced Migraine Prevention Medications: Findings From a Discrete Choice Experiment

Todd Schwedt[1], Ashley Martin[2], Steven Kymes[3], Brian Talon[3], Xin Ying Lee[4], Roger Cady[3,5,6], Divya Asher[3], Meghana Karnik-Henry[3], Emily Mulvihill[2], Dawn Bates[2], Kathleen Beusterien[2], Bernadette Hallissey[2]

[1]Mayo Clinic, Phoenix, AZ, United States; [2]Cerner Enviza, North Kansas City, Kansas City, MO, United States; [3]Lundbeck, LLC, Deerfield, IL, United States; [4]H. Lundbeck A/S, Copenhagen, Denmark; [5]RK Consults, Ozark, MO, United States; [6]Missouri State University, Springfield, MO, United States

Scan the QR code to receive a PDF of the poster

Poster presented at the 27th Annual Meeting of the International Society for Pharmacoeconomics and Outcomes Research
May 15–18, 2022;
National Harbor, Washington, DC

Introduction

- Monoclonal antibodies targeting the calcitonin gene-related peptide (CGRP) pathway are a new class of drugs approved for the preventive treatment of migraine.[1]
 - Oral therapies that have historically been used for migraine prevention, including antidepressants, beta-blockers, and anticonvulsants, have limitations associated with their use, with adverse events and lack of efficacy cited as the most common reasons for discontinuation of treatment.[2]
- The four anti-CGRP monoclonal antibodies for migraine prevention (eptinezumab, erenumab, fremanezumab, and galcanezumab) have demonstrated efficacy and tolerability in randomized, placebo-controlled clinical trials.[3–6] These treatments, as well as onabotulinumtoxinA (indicated for chronic migraine[7]), differ in mode of administration, setting of administration, dosing frequency, and potentially in speed of onset and durability of effect.
- An improved understanding of patient preferences for medication attributes may improve treatment outcomes by helping healthcare providers [HCPs] better match patients with those treatments that best reflect their preferences.

Objectives

- To examine the patient perspective on the importance of treatment attributes, other than efficacy and safety, in the choice of preventive migraine treatment
- To explore potential differences in patient preferences among subgroups determined by latent class analysis

Methods

- This was a non-interventional, cross-sectional study of self-reported preferences for preventive migraine treatment. Eligible patients included adults (≥18 years) in the United States who self-reported a physician diagnosis of migraine, experienced ≥5 monthly migraine days; had taken prescription medication for treatment of migraine; and had tried ≥2 prescription migraine treatments (including current).
- A 25-minute discrete-choice experiment (DCE) online survey was used to assess treatment preferences by having respondents select between two profiles that varied in five attributes: speed of onset (within 24 hours, 1 week, or 3 months), durability of prevention (does not wear off, wears off 1 week prior to next dose, or wears off 2 weeks prior to next dose), mode of administration (IV by HCP, self-injection, or cranial injections by HCP), administration setting (at-home or in-office), and dosing frequency (taken once a month or every 3 months).
 - Combinations of attributes and levels shown in each choice task were based on a balanced overlap design, minimizing potential bias (**Figure 1**).
- Preference weights for each of the DCE attribute levels were estimated using hierarchical Bayesian estimation.
- To compute relative attribute importance at the individual response level, the range of each attribute (the preference weight of most favorable level minus the preference weight of the least favorable level) was divided by the sum of ranges of all attributes and multiplying by 100; thus, the relative importance estimates across all attributes add to 100% per group.
- A latent class analysis (LCA) of the DCE using multinomial logistic regression was used to identify segments of participants that had differing distributions in their preferences.

Results

- A total of 604 respondents were enrolled. The population was primarily white (85.8%) and female (81.5%), with a mean age of 45.9 years.
- In the total migraine sample, all patients were more likely to select the most desirable attribute (e.g., faster speed of onset, longer durability) over less desirable attributes. Respondents most preferred self-injection (over HCP managed care) and preferred at-home administration (over HCP office care), indicating a preference towards self-management of migraine (**Figure 2**).
- In the total migraine sample, mode of administration, durability of prevention, and speed of onset had the highest relative importance, reflecting how much each attribute accounts for variation in preferences (**Table 1**).
 - These attributes had the largest differences in preference weights between the most and least preferred attribute levels.
- While aggregate analyses of the total sample confirmed that mode of administration, durability of prevention, and speed of onset had the highest relative importance, latent class analyses revealed that these attributes were differentially important to certain segments of the migraine population (**Figure 3**):
 - Group 1 (n=128, 22%) favored a self-injection administration and longer durability (i.e., longer intervals between administration periods).
 - Group 2 (n=189, 31%) expressed an aversion to cranial injections (onabotulinumtoxinA).
 - Group 3 (n=158, 26%) prioritized therapies with a more rapid speed of onset.
 - Group 4 (n=129, 21%) favored longer durability with support from their HCP in administration.
- Respondents who preferred self-injection (Group 1) showed the greatest preference for at-home administration among all the LCA groups, whereas respondents who preferred administration by an HCP (Group 4) showed the least preference or a slight avoidance of at-home administration (0.35 vs. -0.22, respectively).
- In **Figure 3** we illustrate the attributes that define the preference structure of the four groups identified in latent class analysis. Each axis illustrates the difference between the highest and lowest level of the attribute in that subgroup. Thus, a higher score on the axis indicates a greater importance for that attribute in the subgroup. From this, we see that Groups 2 and 3 have clearly defining attribute. For Group 2 it is a preference to avoid cranial injections and in Group 3, it is the speed of onset (with durability of effect as a secondary factor). Groups 1 and 4 have a more balanced preference structure, lacking as strong a single determining attribute. In Group 1, the strongest attribute is Mode of Administration, with the respondents indicating a preference for self-administration, but none of the attributes have a utility difference greater than 1. In Group 4 there is also no utility difference greater than 1, but in this group longer durability is preferred, with mode of administration and setting indicating a preference for HCP based administration.

Figure 1. Example of a Single Discrete-Choice Experiment Choice Task

Assuming everything else to be the same between the options below, which would you most prefer?

Option 1	Option 2
After taking for the first time, wait 3 months before fully effective in preventing migraines	After taking for the first time, wait 1 week before fully effective in preventing migraines
Taken once every 3 months	Taken once a month
Starts to wear off 2 weeks before next dose	Starts to wear off 2 weeks before next dose
Self-inject treatment under the skin with an auto-injector	Healthcare provider administers 30 Botox injections across face/neck
Administered at home	Administered at a doctor's office or infusion center
Select	Select

Botox® was included by brand name given that it is the only botulinum toxin A approved for treatment of (chronic) migraine and is a recognizable proprietary eponym.

Figure 2. Attribute-Level Preference Weights in the Total Migraine Sample

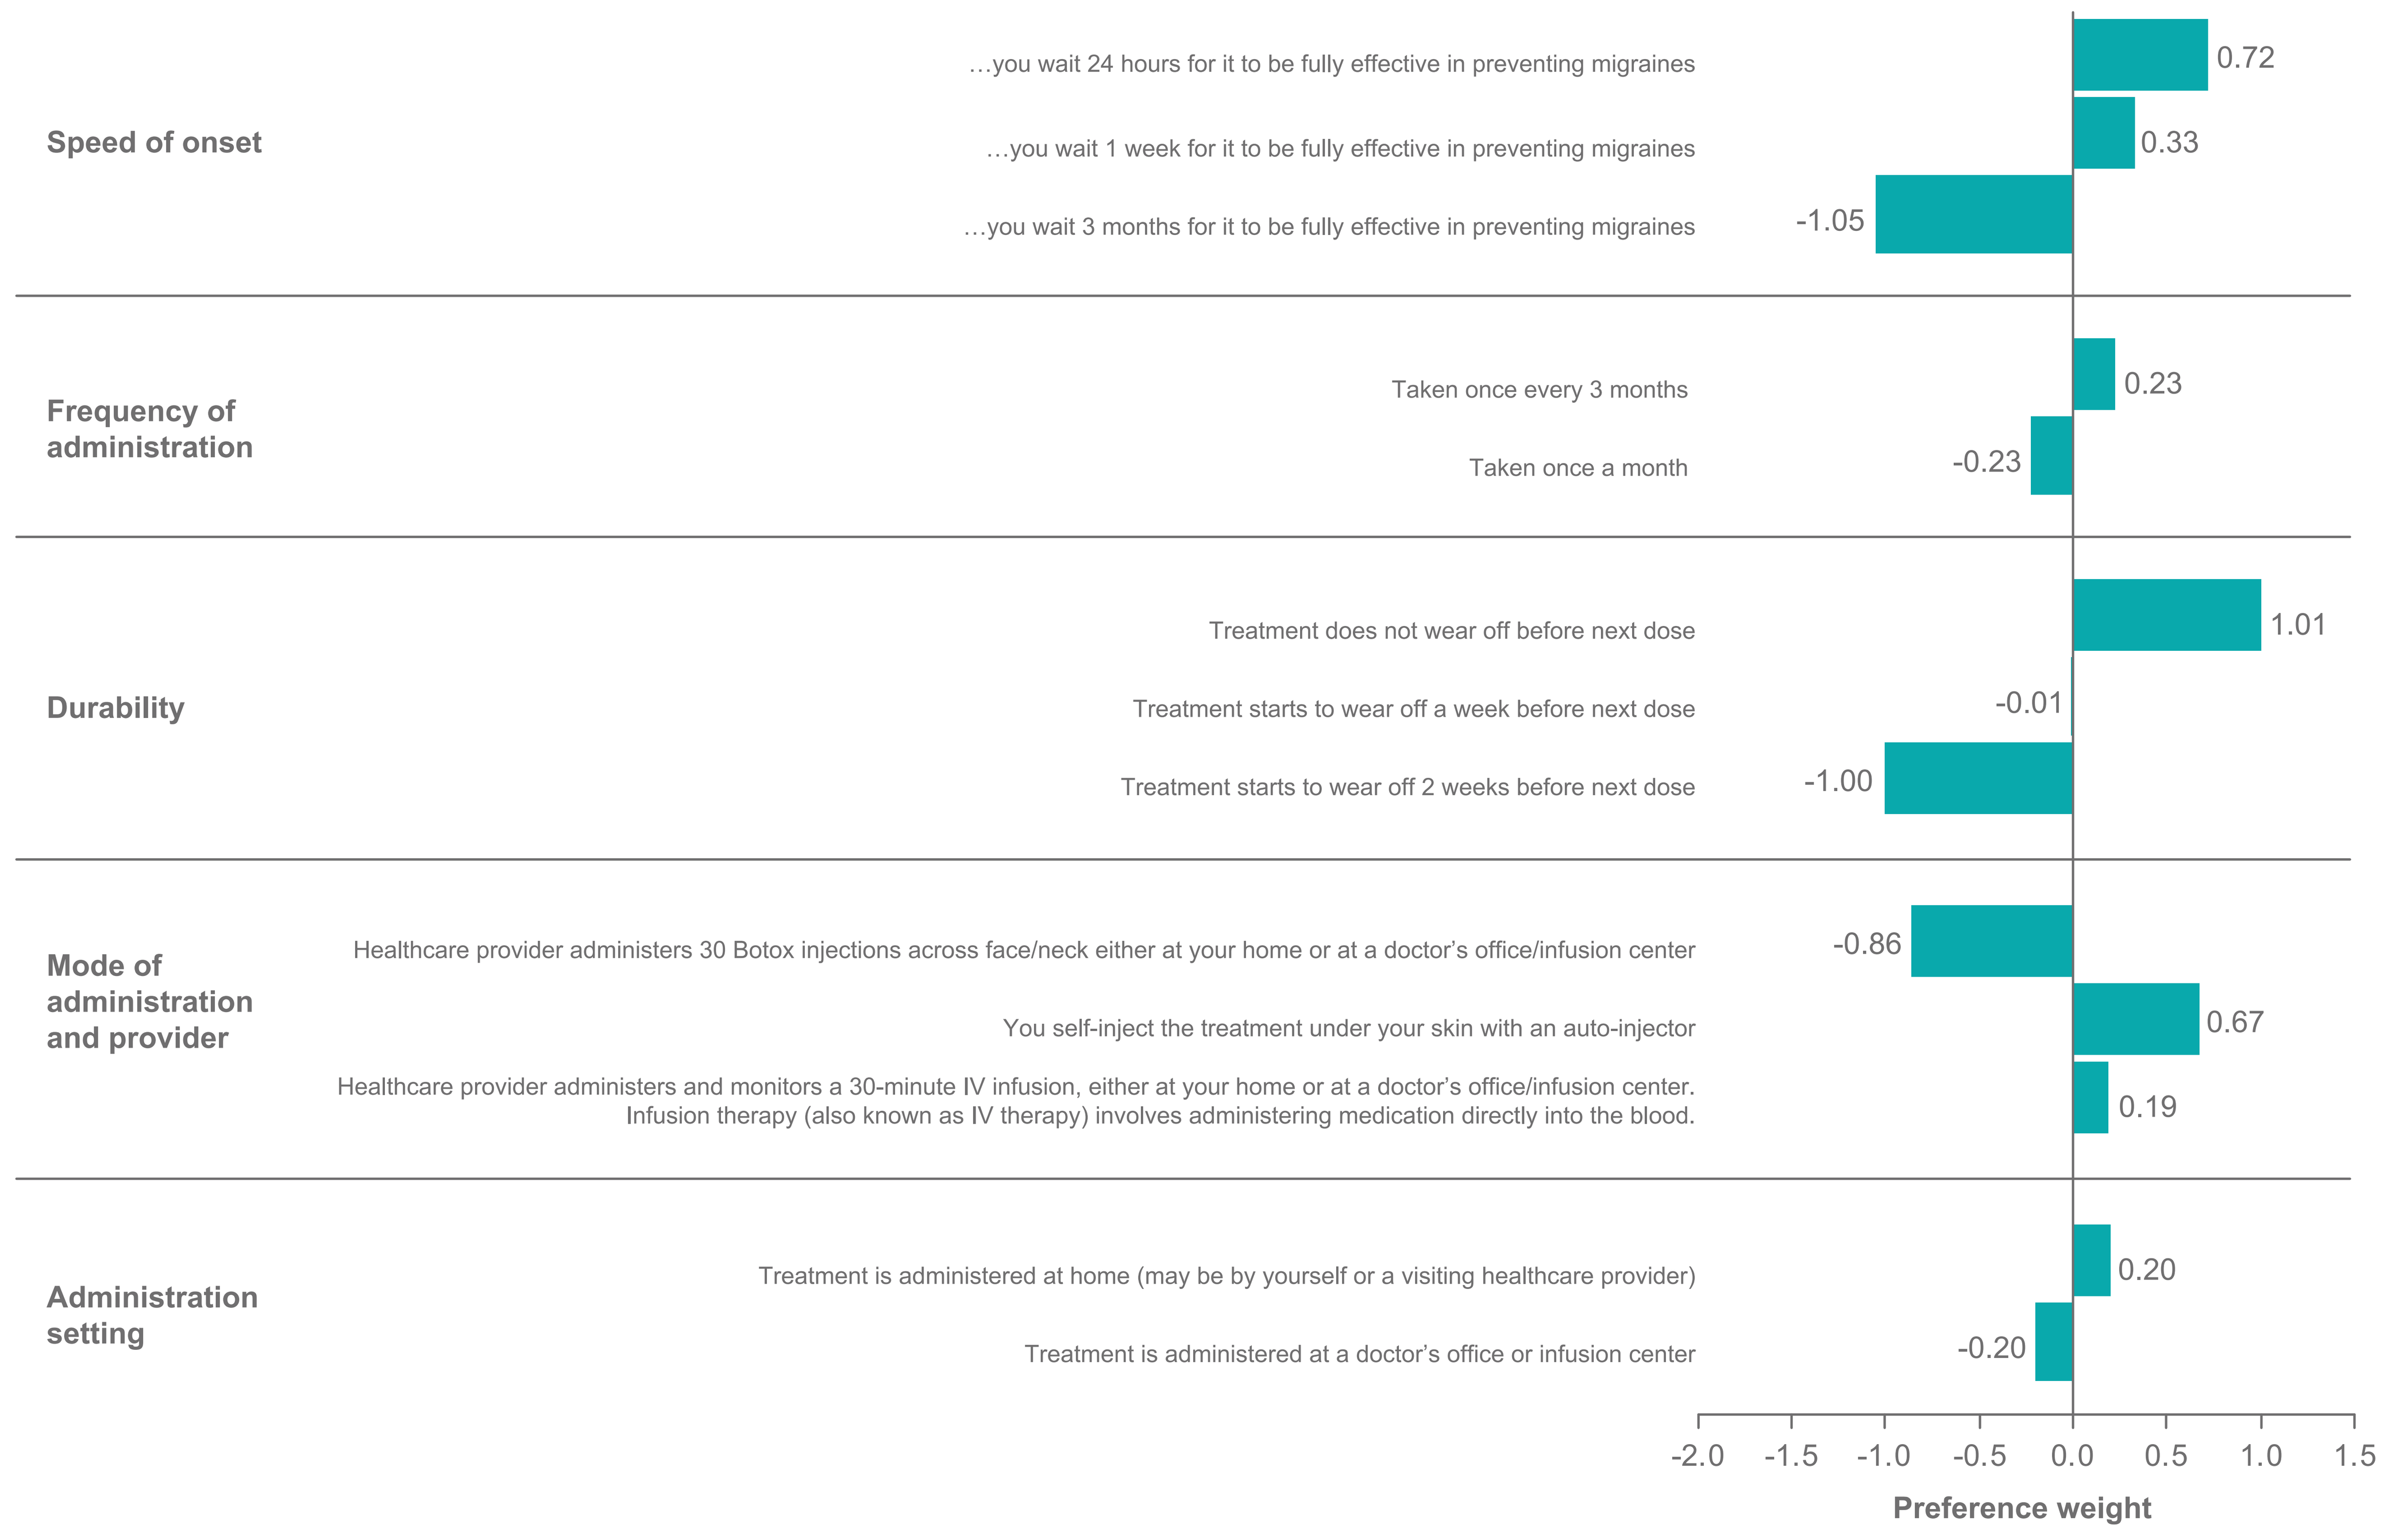

Speed of onset
…you wait 24 hours for it to be fully effective in preventing migraines
0.72
…you wait 1 week for it to be fully effective in preventing migraines
0.33
…you wait 3 months for it to be fully effective in preventing migraines
-1.05
Frequency of administration
Taken once every 3 months
0.23
Taken once a month
-0.23
Durability
Treatment does not wear off before next dose
1.01
Treatment starts to wear off a week before next dose
-0.01
Treatment starts to wear off 2 weeks before next dose
-1.00
Mode of administration and provider
Healthcare provider administers 30 Botox injections across face/neck either at your home or at a doctor's office/infusion center
-0.86
You self-inject the treatment under your skin with an auto-injector
0.67
Healthcare provider administers and monitors a 30-minute IV infusion, either at your home or at a doctor's office/infusion center. Infusion therapy (also known as IV therapy) involves administering medication directly into the blood.
0.19
Administration setting
Treatment is administered at home (may be by yourself or a visiting healthcare provider)
0.20
Treatment is administered at a doctor's office or infusion center
-0.20
-2.0
-1.5
-1.0
-0.5
0.0
0.5
1.0
1.5
Preference weight

Preference weights should not be interpreted by themselves. Instead, the magnitude of change within one attribute should be compared to change within another attribute.

Table 1. Attribute Relative Importance in the Total Migraine Sample and by LCA Subgroups (Mean Percentage)

		Subgroups With Differing Preferences			
	Total Migraine Sample (N=604)	Group 1: Auto-Inject & Longer Durability (n=128)	Group 2: Averse to Cranial Injections (n=189)	Group 3: Faster Speed of Onset (n=158)	Group 4: IV Infusion & Longer Durability (n=129)
Mode of administration and provider	**28.8%**	**27.3%**	**51.9%**	7.8%	**26.0%**
Durability of effect	**27.0%**	**22.7%**	**19.9%**	**34.2%**	**41.1%**
Speed of onset	25.5%	14.2%	13.6%	**53.4%**	12.9%
Administration setting	9.9%	20.0%	6.9%	3.3%	19.3%
Frequency of administration	8.8%	15.9%	7.7%	1.3%	0.7%

Each column sums to 100. Percentages represent the mean % variation in preferences explained by the attribute within that sample/subgroup. IV, intravenous; LCA, latent class analysis.

Figure 3. Largest Difference in Attribute Preference Weights by LCA Subgroups

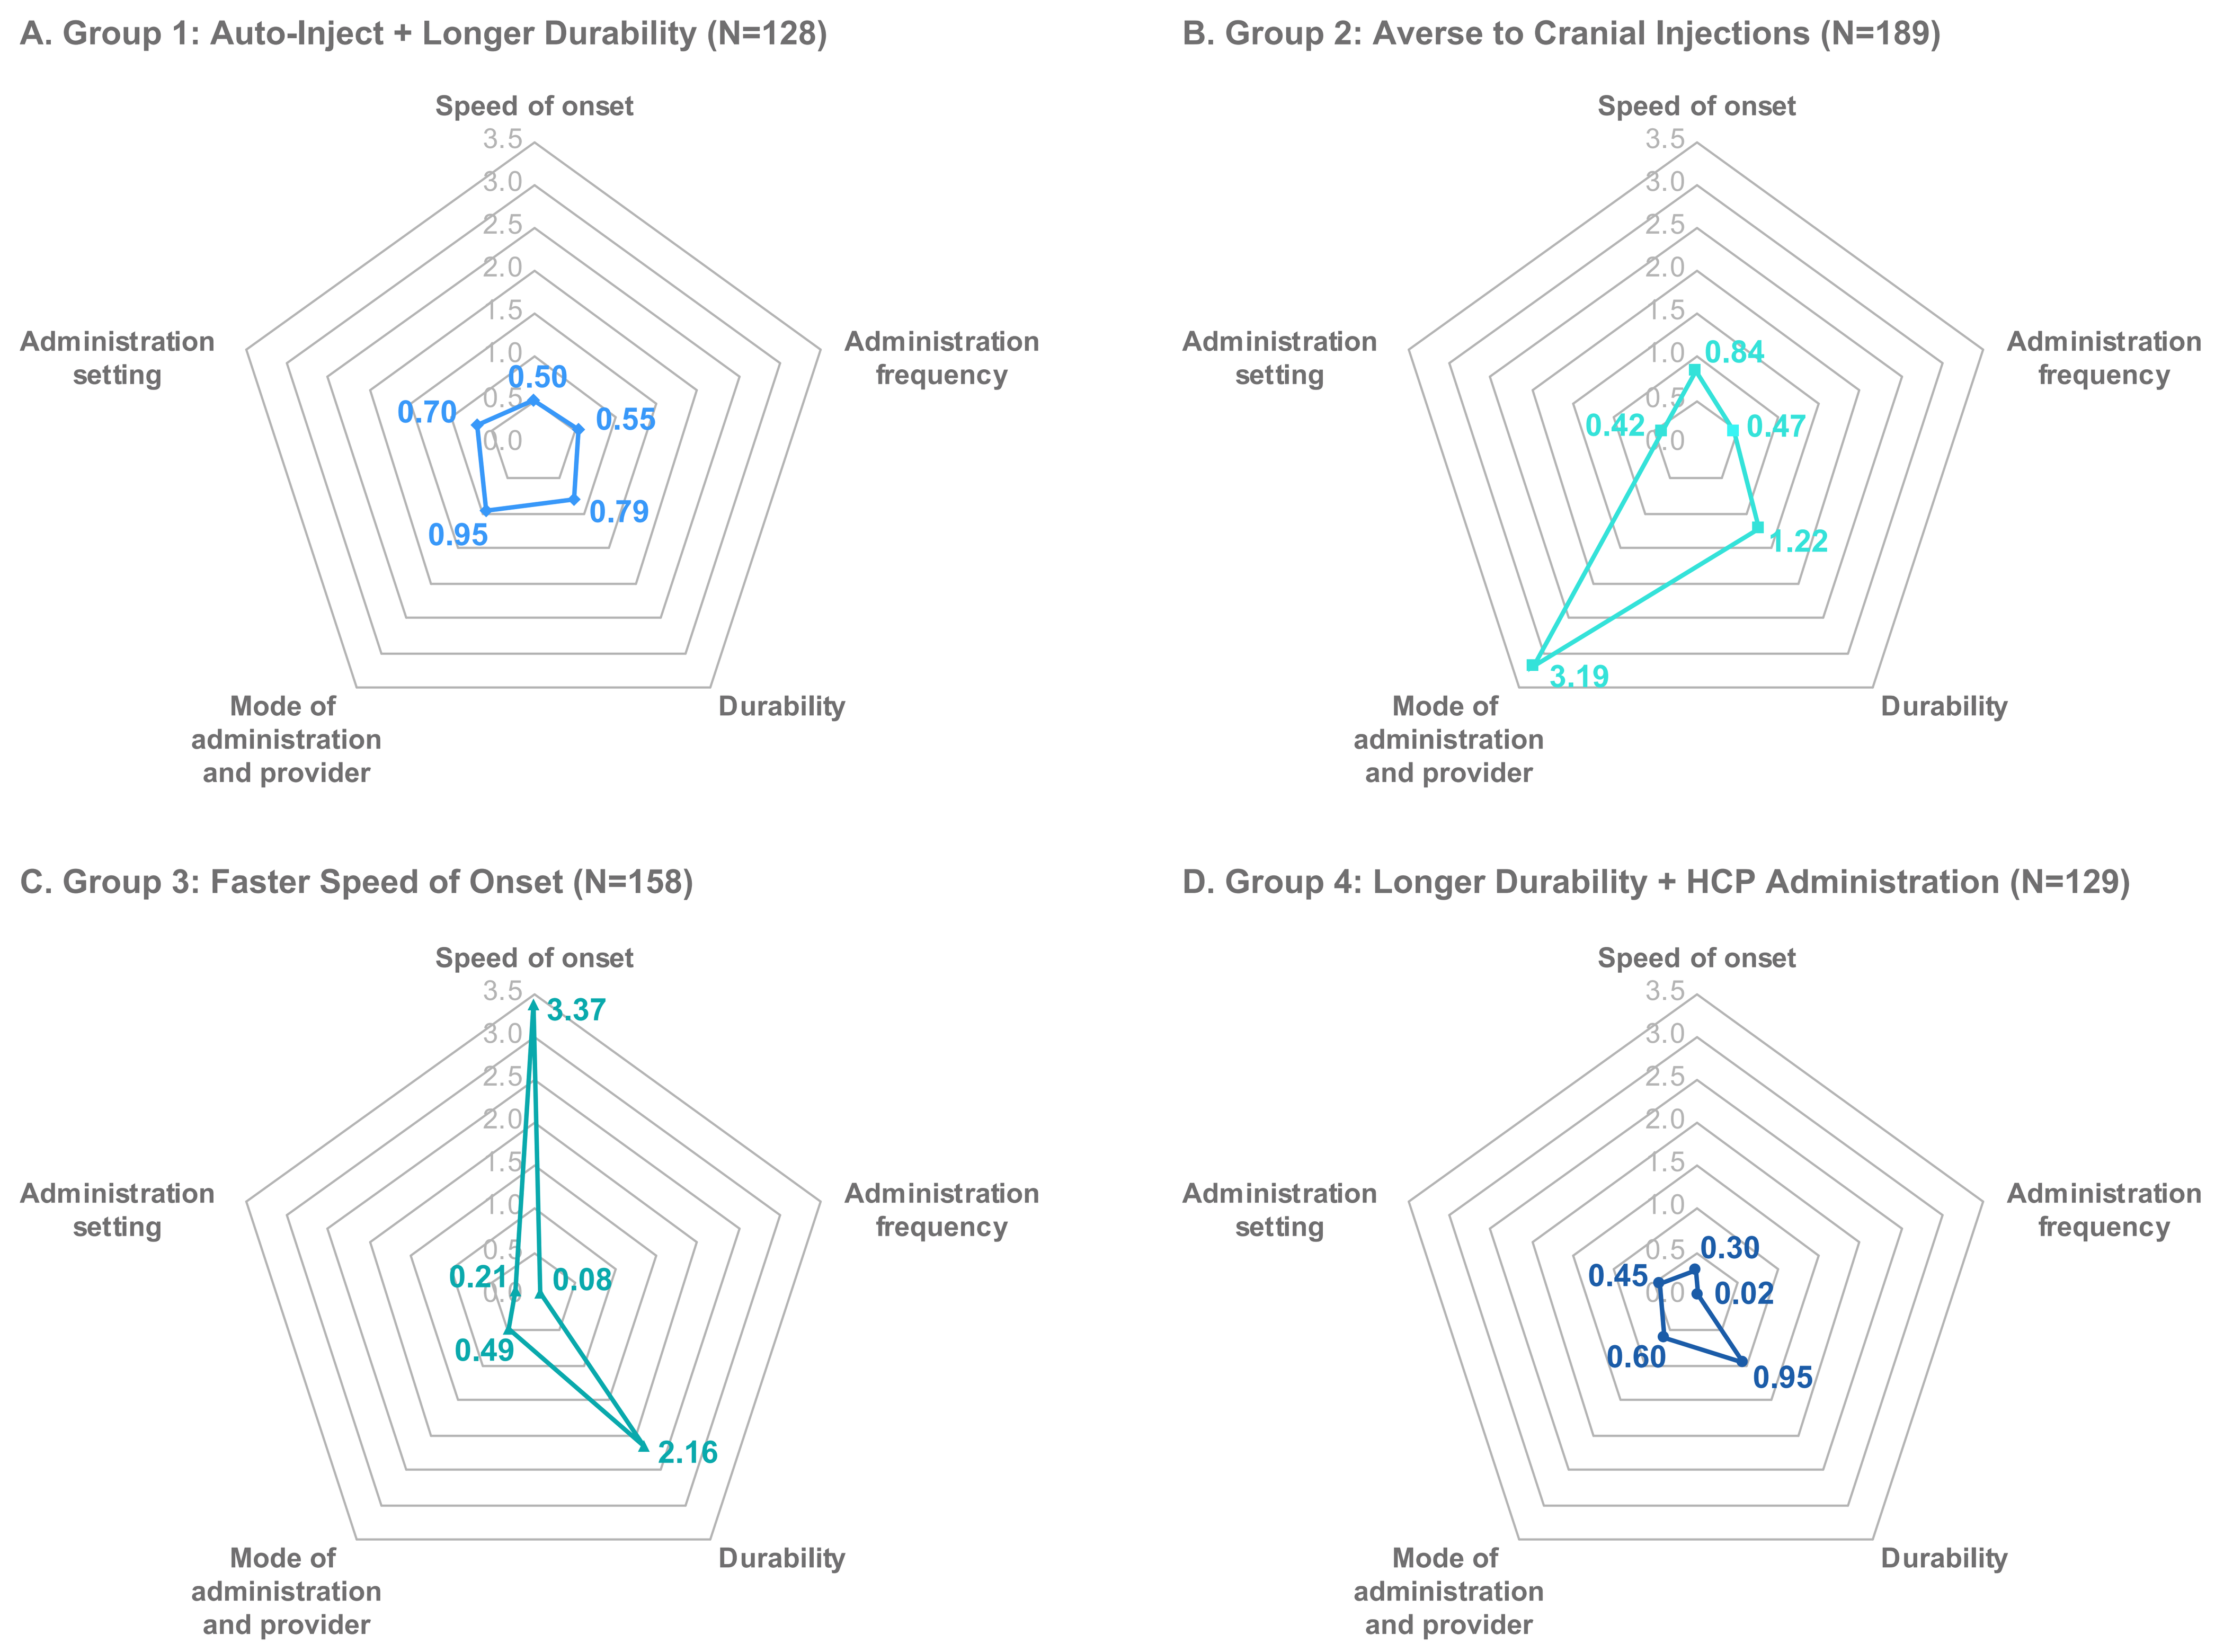

A. Group 1: Auto-Inject + Longer Durability (N=128)
Speed of onset
Administration frequency
Durability
Mode of administration and provider
Administration setting
0.50
0.55
0.79
0.95
0.70
B. Group 2: Averse to Cranial Injections (N=189)
0.84
0.47
1.22
3.19
0.42
C. Group 3: Faster Speed of Onset (N=158)
3.37
0.08
2.16
0.49
0.21
D. Group 4: Longer Durability + HCP Administration (N=129)
0.30
0.02
0.95
0.60
0.45
0.0
0.5
1.0
1.5
2.0
2.5
3.0
3.5

Values represent the absolute difference between the most (highest) and least (lowest) preferred level within each attribute HCP, healthcare provider.

KEY POINTS

- When considering preventive migraine treatment, patients placed greatest importance on mode of administration, durability of prevention, and speed of onset; administration setting and frequency had a lower relative importance.
- Latent class analyses revealed subgroups that differ in the strength of their preference for mode of administration, durability of prevention, and speed of onset.
- Three-fourths of participants were not averse to IV infusion as a route of administration. However, attribute preferences should not be considered individually; the treatment plan will depend on the total assessment of the benefits.

CONCLUSIONS

- Together, these results highlight mode of administration, durability of prevention, and speed of onset as key factors in treatment choice among US adults with migraine, with latent class analyses revealing subgroups that differ in their treatment preferences.
- HCPs who treat patients with migraine should consider that their patients may vary with respect to their treatment preferences.
- Given the varied attributes of available treatment for migraine prevention and the impact of patient satisfaction on outcomes, these results underscore the need for individualized treatment that accounts for patient preferences of treatment.

References

1. Raffaelli B, et al. *Expert Opin Biol Ther.* 2019;19:1307-1317.
2. Moriarty M, et al. *J Nurse Practitioners.* 2019;15(10):717-724.e1.
3. Vyepti. Package insert. Lundbeck Seattle BioPharmaceuticals, Inc.; 2021.
4. Aimovig. Package insert. Amgen Inc.; 2021.
5. Ajovy. Package insert. Teva Pharmaceuticals USA, Inc.; 2021.
6. Emgality. Package insert. Eli Lilly and Company; 2021.
7. Botox. Package insert. Allergan; 2021.

Disclosures

TS has received consulting fees from AbbVie, Alder/Lundbeck, Allergan, Biodelivery Science, Biohaven, Cipla, Click Therapeutics, Eli Lilly, Equinox, Ipsen, Novartis, Tonix, Weber and Weber, and XoC; is or was a board member of American Headache Society, American Migraine Foundation, International Concussion Society, and International Headache Society (past); holds stocks or stock options in Aural Analytics and Nocira; has received grants or contracts from Amgen, Patient Centered Outcomes Research Institute, National Institutes of Health, United States Department of Defense, Henry Jackson Foundation, and Mayo Clinic; has received publishing royalties from Up To Date; and has a patent for "Speech Analysis Devices and Methods for Identifying Migraine Attacks" (Application #20220005494). **AM, EM, DB,** and **KB** are employees of Cerner Enviza, a company that received funding from Lundbeck LLC for time spent conducting this research. **SK, BT, XYL, DA,** and **MKH** are employees of Lundbeck LLC. **RC** was an employee of Lundbeck LLC at the time of study completion. **BH** was an employee of Cerner Enviza, a company that received funding from Lundbeck LLC for time spent conducting this research, at the time of study completion.

Acknowledgements

This study was sponsored by Lundbeck LLC (Deerfield, IL, United States). The authors thank The Medicine Group, LLC (New Hope, PA, United States) for providing medical writing support, which was funded by Lundbeck LLC and in accordance with Good Publication Practice guidelines.