Potential Budget Impact of Finerenone in Patients with Chronic Kidney Disease and Type 2 Diabetes Being Treated with Standard of Care

Victoria W Dayer[1], Ryan N Hansen[1], Rakesh Singh[2], Sheldon X Kong[2], Todd Williamson[2], Sean D Sullivan[1]
[1]University of Washington, Seattle, WA, USA, [2]Bayer U.S. LLC, Whippany, NJ, USA

UNIVERSITY of WASHINGTON
THE CHOICE INSTITUTE
School of Pharmacy

OBJECTIVE

Finerenone is a nonsteroidal, selective mineralocorticoid receptor antagonist indicated in adult patients with chronic kidney disease (CKD) and type 2 diabetes (T2D).[1]

We estimated the 3-year financial impact of adding finerenone to United States health plan formularies, as an add-on to standard of care.

KEY MODEL ASSUMPTIONS & LIMITATIONS

> Hypothetical 1,000,000 member health plan with Commercial or Medicaid lives

> Payer perspective

> Time horizon: 3 years

> Standard of care drug costs are assumed similar in both groups and are therefore omitted

> Plan population size is constant

> Uptake was projected to be 1.2%, 4.3%, and 6.6% in eligible patients in years 1, 2, and 3, respectively.[2]

> Due to the lack of a direct comparator other than standard of care at the time of the development of this model, this model does not account for any effect that finerenone may have on the use and market share of other treatment options, such as SGLT-2 inhibitors.

> Mortality and disease incidence are assumed to be accounted for by disease prevalence estimates

POPULATION

The population was selected based on the patient populations in the Finerenone CKD Clinical Trial Programs (FIGARO and FIDELIO).[3-4]

> Commercial and Medicare scenarios were modeled separately

TABLE 1. EPIDEMIOLOGY INPUTS

Total members in plan	1,000,000
Prevalence of T2D[5-6]	10.5%
Prevalence of CKD in T2D[5,7-8]	39%
Proportion w/ albuminuria[6,9-10]	53%
Diagnosed CKD[6,9-10]	47%
Treated (SoC) CKD[6,10]	83%
Eligible (exclude HFrEF)[6,10]	94%

FIGURE 1. PATIENT POPULATION

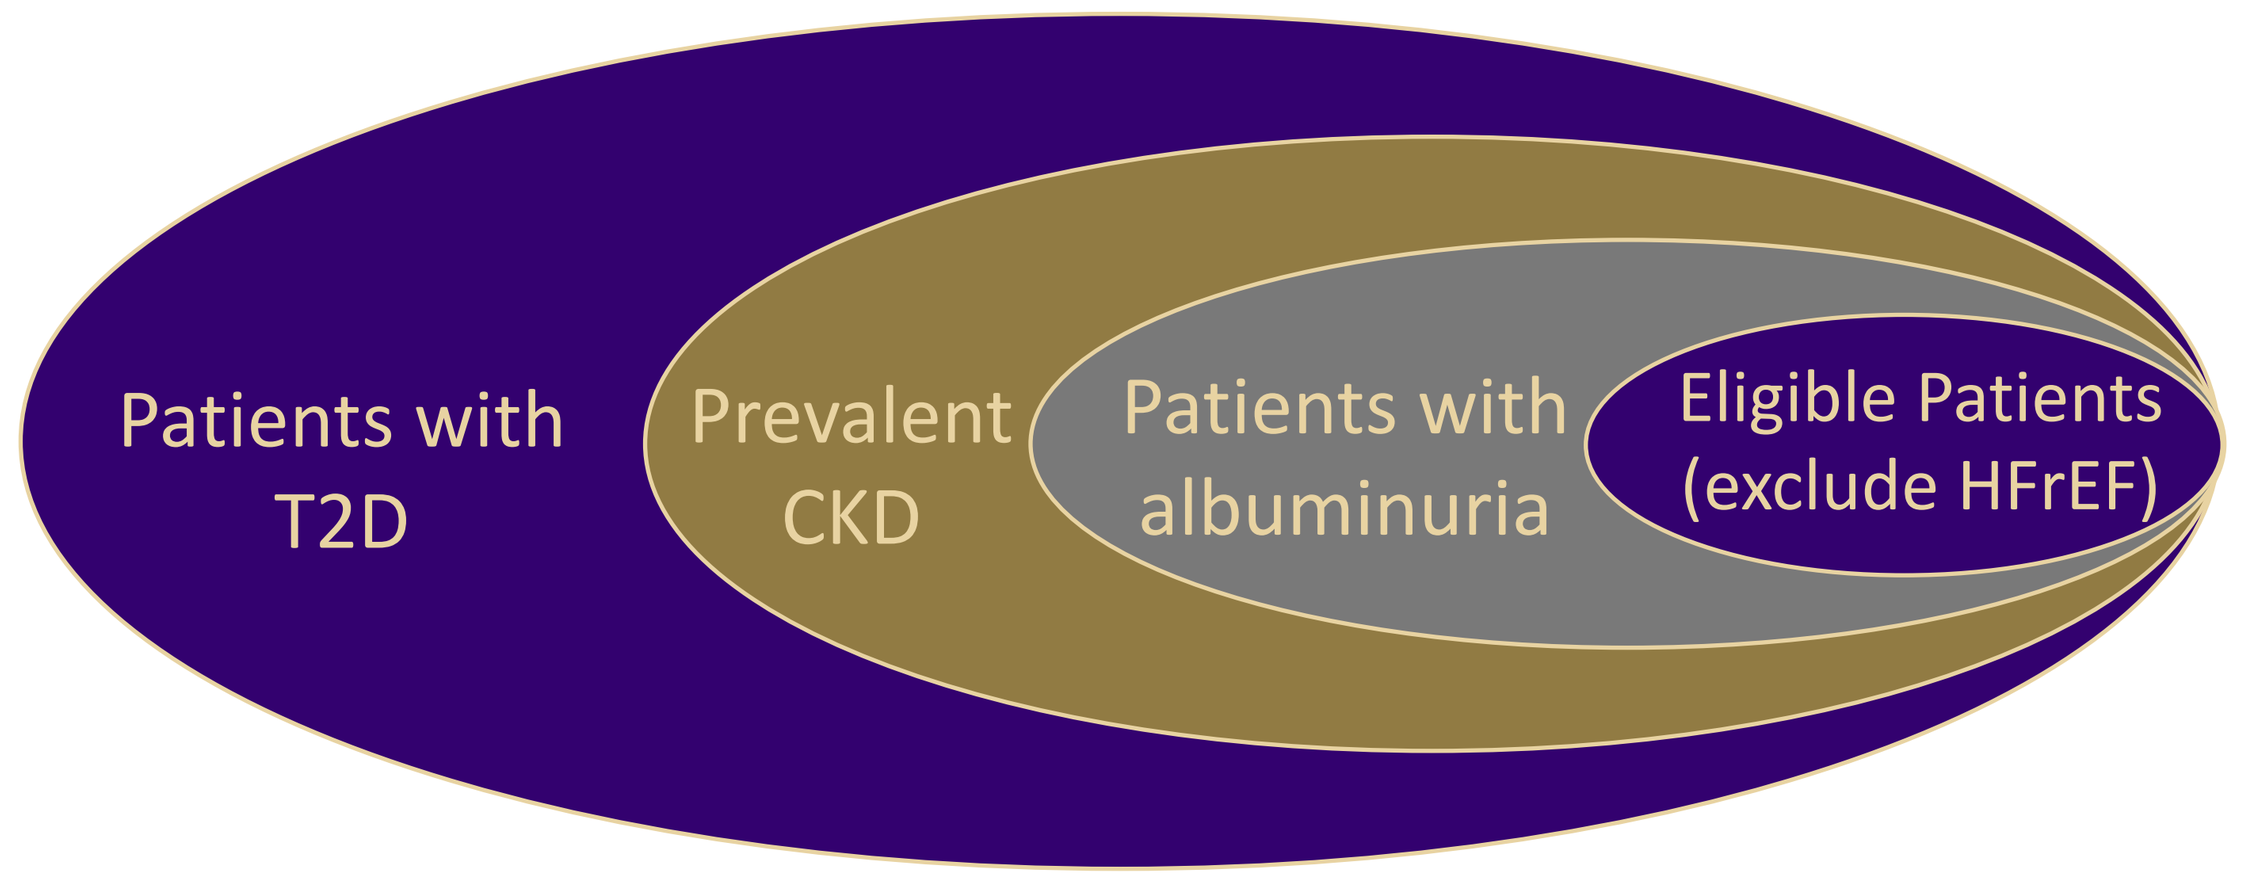
Patients with T2D
Prevalent CKD
Patients with albuminuria
Eligible Patients (exclude HFrEF)

COSTS & COST-SHARING

> **Drug costs:** A price of $18.97/tablet was used for finerenone.[1,11] Standard of care drug costs are omitted.

> **Medical costs**: Differences in event rates for renal and CV outcomes are included with their associated medical costs and AE/Other health event cost offsets, derived from the finerenone US Cost-Effectiveness Analysis.[5,12-15]

TABLE 2. MEDICAL COSTS

Medical Costs[15]	Cost (per patient)
Finerenone group*: Mean annual medical cost	$13,488
Finerenone group*: AE/Other health events	$728
SoC group: Mean annual medical cost	$13,562
SoC group: AE/Other health events	$740

*Treated with finerenone added to standard of care

> **Patient cost-sharing:**

TABLE 3. COST-SHARING

Cost-sharing model	Value	% of patients
Commercial: copay	$50	80%
Commercial: coinsurance	15%	20%
Medicare: coinsurance	5%	100%

RESULTS

Addition of finerenone to the formulary of this hypothetical United States health plan resulted in moderately increased costs over the course of 3 years.

> Minor cost savings due to renal and cardiovascular outcomes averted compared to standard of care alone.

> Medicare population budget impact was higher due to the larger population size and differences in patient cost-sharing.

> Commercial Incremental Budget Impact (3 year): $5,927,000

> Medicare Incremental Budget Impact (3 year): $6,256,000

FIGURE 2. INCREMENTAL BUDGET IMPACT

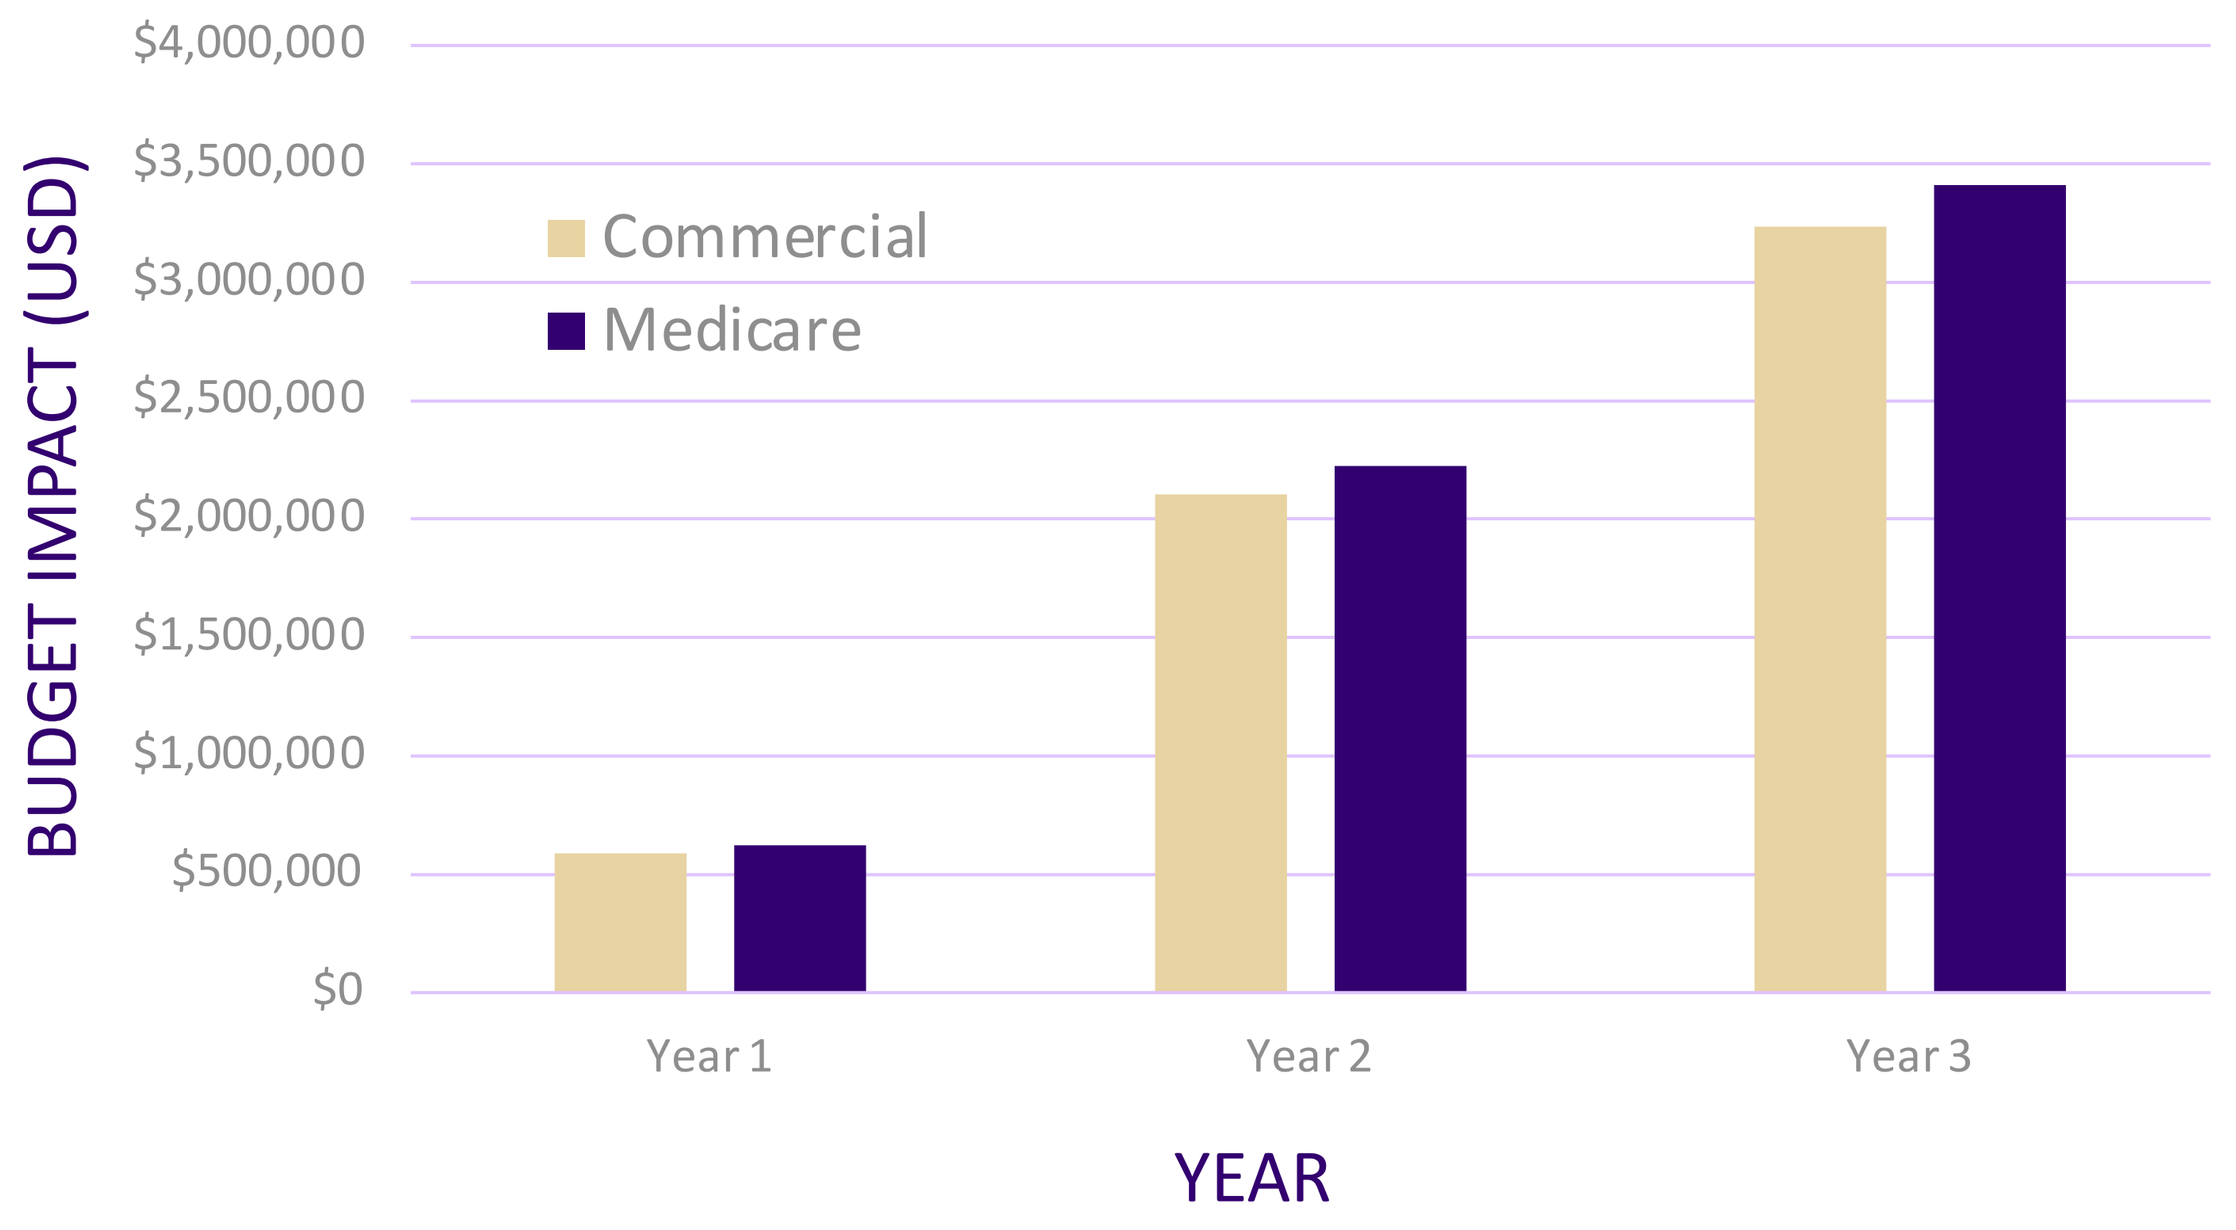
$4,000,000
$3,500,000
$3,000,000
$2,500,000
$2,000,000
$1,500,000
$1,000,000
$500,000
$0
BUDGET IMPACT (USD)
Commercial
Medicare
Year 1
Year 2
Year 3
YEAR

FIGURE 3. ONE-WAY SENSITIVITY ANALYSIS (MEDICARE)

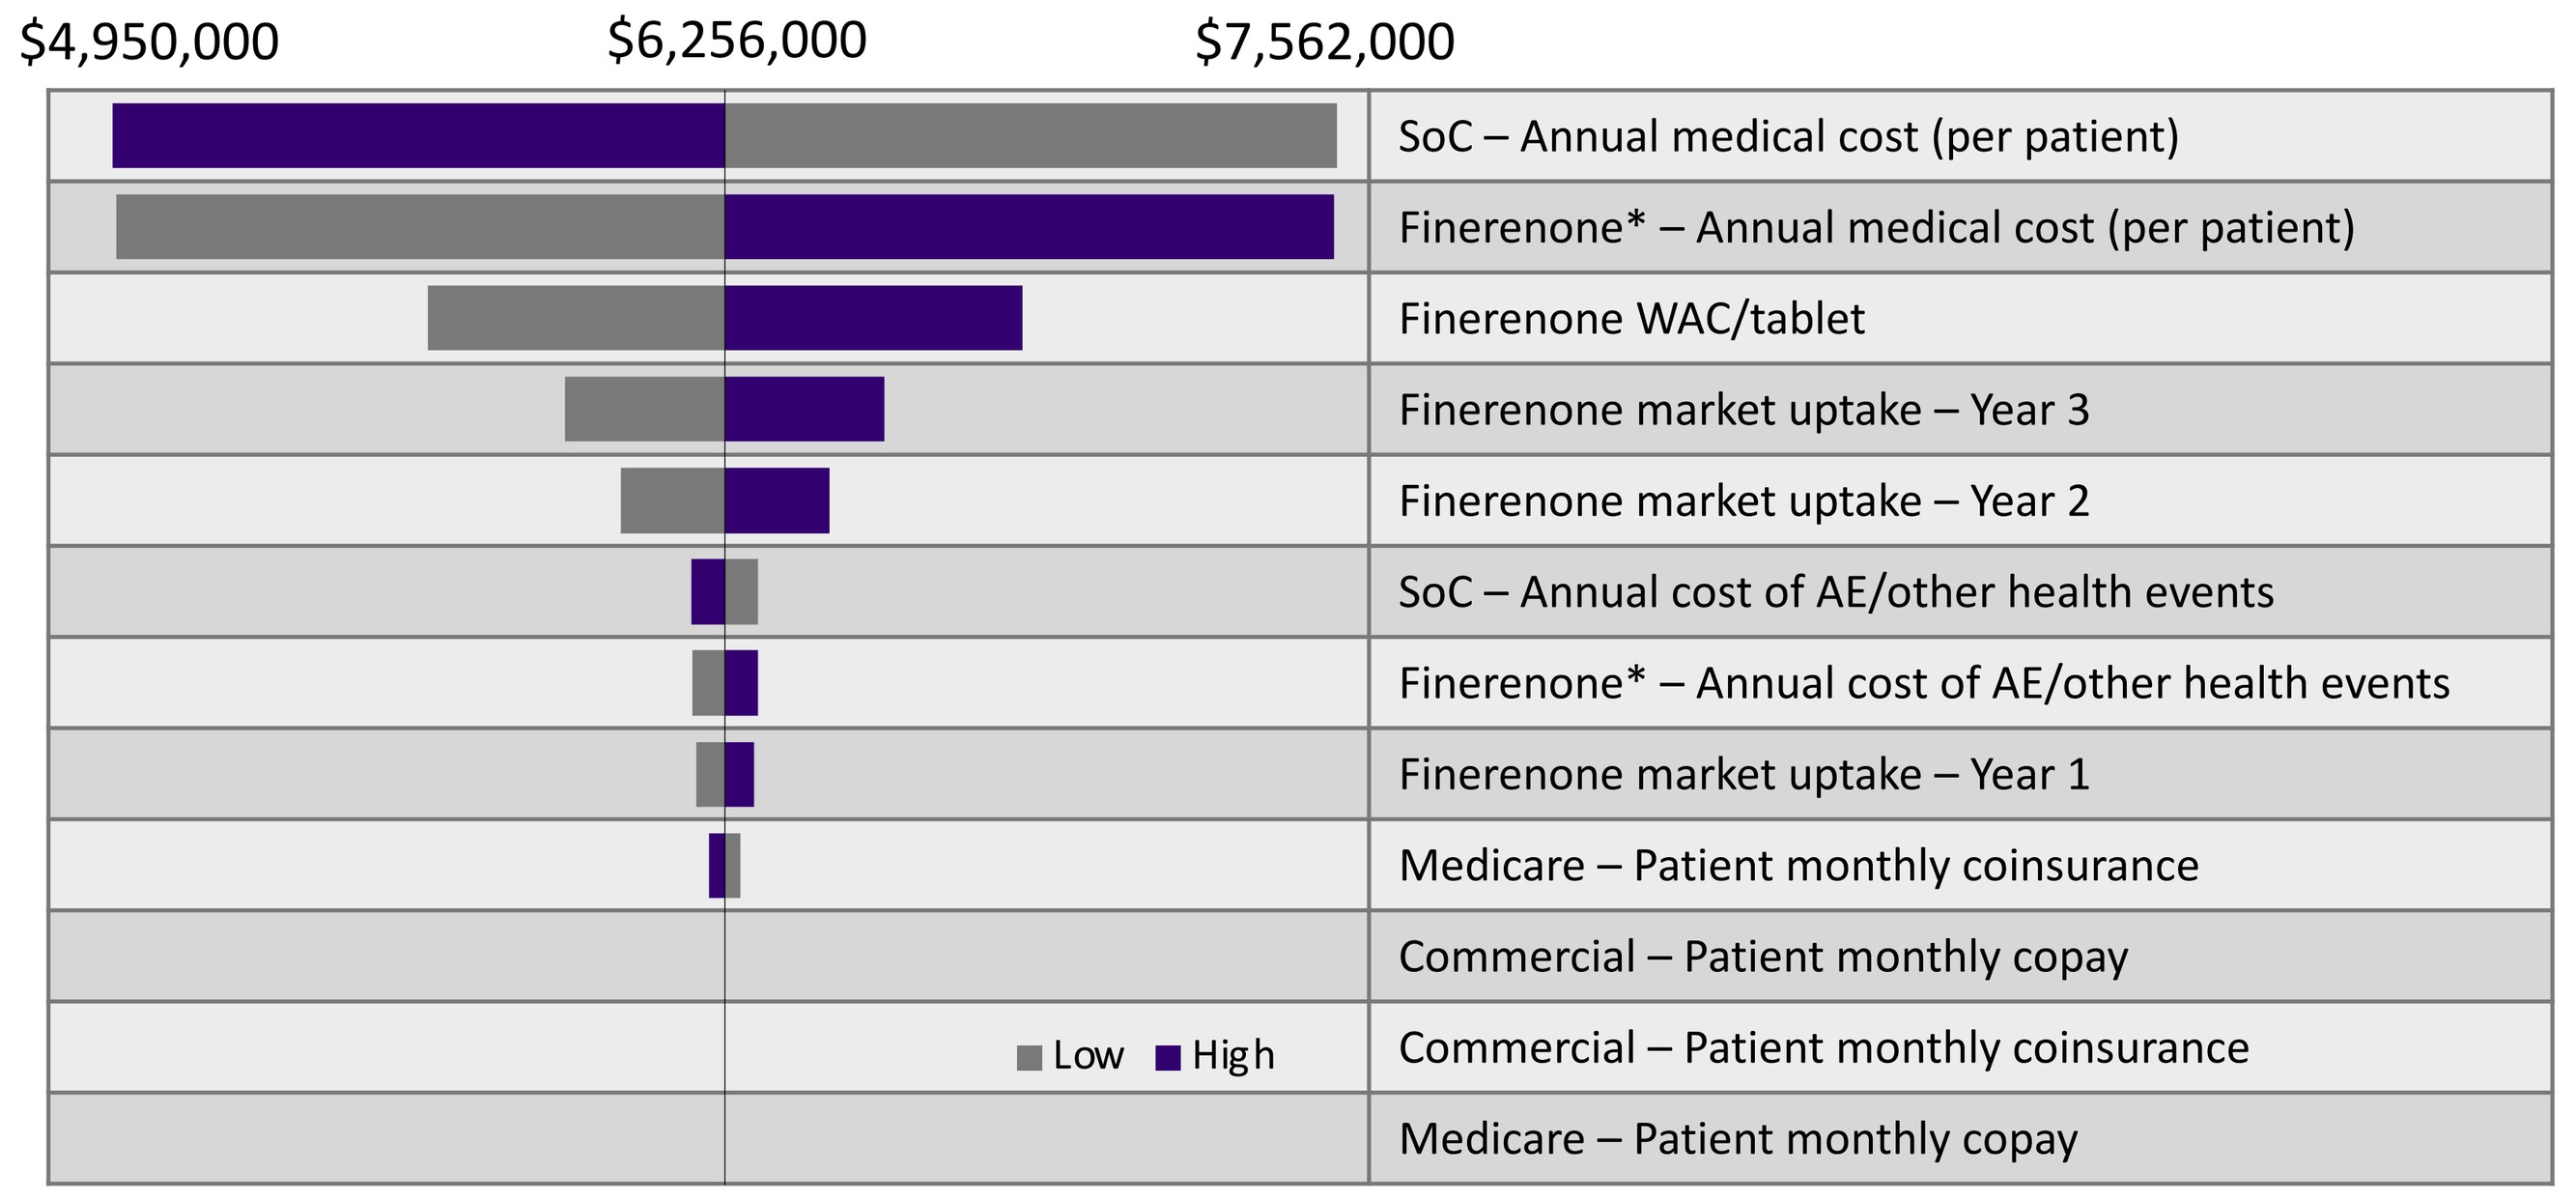
$4,950,000
$6,256,000
$7,562,000
SoC – Annual medical cost (per patient)
Finerenone* – Annual medical cost (per patient)
Finerenone WAC/tablet
Finerenone market uptake – Year 3
Finerenone market uptake – Year 2
SoC – Annual cost of AE/other health events
Finerenone* – Annual cost of AE/other health events
Finerenone market uptake – Year 1
Medicare – Patient monthly coinsurance
Commercial – Patient monthly copay
Commercial – Patient monthly coinsurance
Medicare – Patient monthly copay
Low
High

*Treated with finerenone added to standard of care
OWSA results for the Commercial scenario were consistent

TABLE 4. INCREMENTAL PMPM IMPACT

Commercial	Year 1	Year 2	Year 3
Incremental PMPM - pharmacy	$0.05	$0.18	$0.27
Incremental PMPM - medical	$0.00	$0.00	$0.00
Incremental PMPM - total	$0.05	$0.18	$0.27
Medicare	**Year 1**	**Year 2**	**Year 3**
Incremental PMPM - pharmacy	$0.05	$0.19	$0.29
Incremental PMPM - medical	$0.00	$0.00	$0.00
Incremental PMPM - total	$0.05	$0.19	$0.28

PMPM: per-member-per-month

CONCLUSIONS

> Adding finerenone to the formulary of a hypothetical 1,000,000 member US health plan as an add-on to standard of care resulted in an expected budget impact of approximately $5,927,000 in a Commercial population and $6,256,000 in a Medicare population over a 3-year time horizon.

> Incremental budget impact was primarily attributable to finerenone costs, with minor cost offsets due to medical care savings.

> Incremental per-member-per-month costs would be expected to increase year-over-year due to increased uptake.

> Medical cost impacts and market uptake are key drivers of the model results as identified by the one-way sensitivity analysis.

REFERENCES

1. KERENDIA Prescribing Information. Bayer HealthCare Pharmaceuticals, Inc. July 2021.; 2. Kerendia's TRx uptake compared to analogues By Scenario, Bayer Data on File; 3. Bakris GL, Agarwal R, Anker SD, et al. Effect of Finerenone on Chronic Kidney Disease Outcomes in Type 2 Diabetes. N Engl J Med. 2020;383(23):2219-2229. doi:10.1056/NEJMoa2025845; 4. Pitt B, Filippatos G, Agarwal R, Anker SD, Bakris GL, Rossing P, Joseph A, Kolkhof P, Nowack C, Schloemer P, Ruilope LM; FIGARO-DKD Investigators. Cardiovascular Events with Finerenone in Kidney Disease and Type 2 Diabetes. N Engl J Med. 2021 Dec 9;385(24):2252-2263. doi: 10.1056/NEJMoa2110956. Epub 2021 Aug 28. PMID: 34449181.; 5. National Diabetes Statistics Report. Published online 2020. https://www.cdc.gov/diabetes/pdfs/data/statistics/national-diabetes-statistics-report.pdf; 6. Finerenone Forecast Final Assumptions and Outputs, Bayer Data on File.; 7. Duru OK, Middleton T, Tewari MK, Norris K. The Landscape of Diabetic Kidney Disease in the United States. Curr Diab Rep. 2018;18(3):14. Published 2018 Feb 19. doi:10.1007/s11892-018-0980-x; 8. Wu B, Bell K, Stanford A, et al. Understanding CKD among patients with T2DM: prevalence, temporal trends, and treatment patterns-NHANES 2007-2012. BMJ Open Diabetes Res Care. 2016;4(1):e000154. Published 2016 Apr 11. doi:10.1136/bmjdrc-2015-000154; 9. NHANES 1999-2012 Analysis via Tessellon Model, Bayer Data on File.; 10. Epidemiology Worksheet, Bayer Data on File.; 11. KERENDIA 10 mg and 20 mg. Analysource Online. http://www.analysource.com; 12. American Diabetes Association. Statistics about Diabetes. Accessed July 14, 2021. https://www.diabetes.org/resources/statistics/statistics-about-diabetes ; 13. Bailey RA, Wang Y, Zhu V, Rupnow MF. Chronic Kidney Disease in US Adults with Type 2 Diabetes: an Updated National Estimate of Prevalence Based on Kidney Disease: Improving Global Outcomes (KDIGO) Staging. BMC Res Notes. 2014;7(1):415. doi:10.1186/1756-0500-7-415; 14. American Diabetes Association. Standards of Medical Care in Diabetes — 2020. Dia Care. 2020;43(Supplement 1):S7-S13. doi:10.2337/dc20-S001; 15. Finerenone US Cost-Effectiveness Analysis, Bayer Data on File.