Sociodemographics may Influence the Use of Sugammadex vs. Neostigmine for Neuromuscular Blockade Reversal

Tatiana Ferraro, Kunal Lodaya, Jonathan Lilley, Fabian T.W. D'Souza

Boston Strategic Partners, Inc.

BACKGROUND

Sugammadex is a novel neuromuscular blockade (NB) reversal agent that has demonstrated a faster and safer NB reversal than acetylcholinesterase inhibitors such as neostigmine.[1,2] Despite published literature on cost-effectiveness, little is known about the influence of sociodemographic factors on the administration of sugammadex vs. neostigmine[3,4,5]

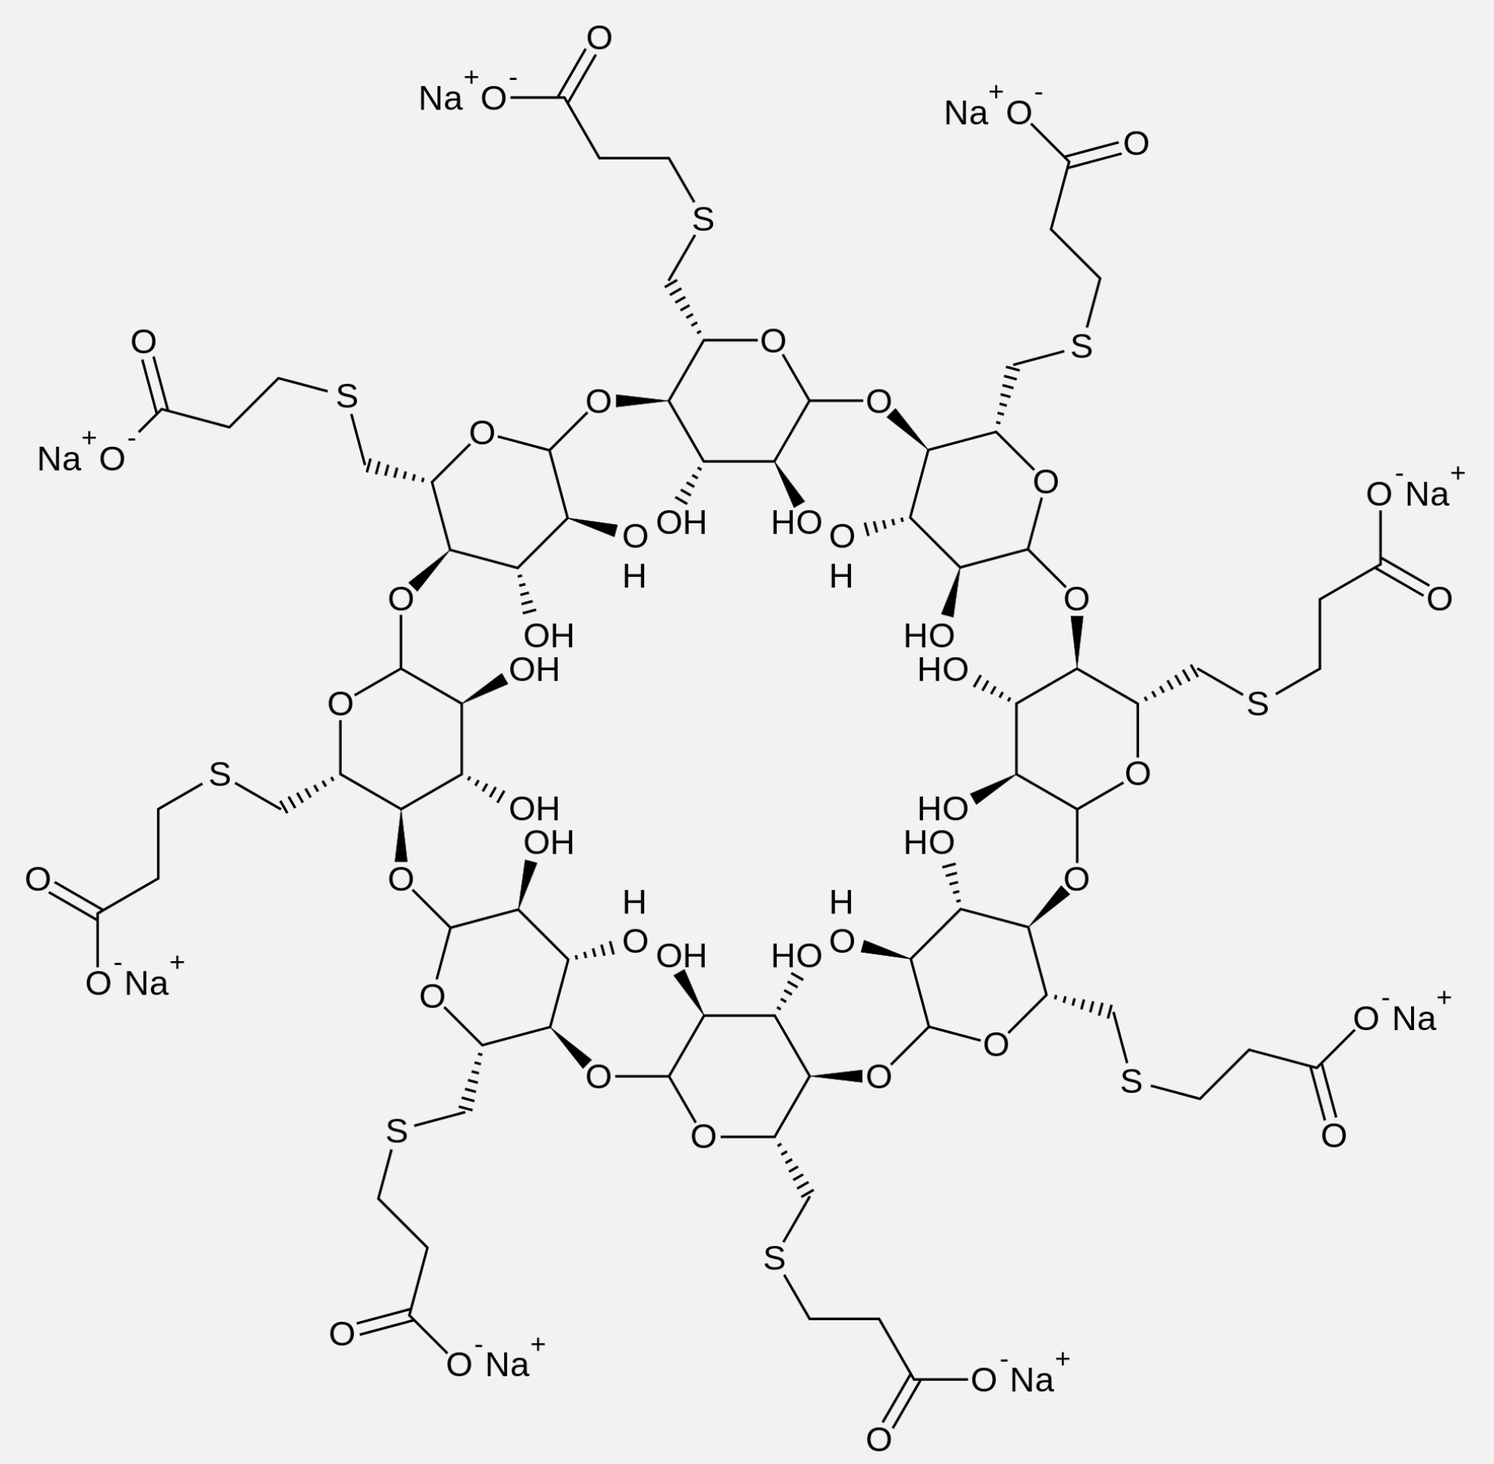

Structural Diagram of Sugammadex Sodium[6]

OBJECTIVES

To understand potential demographic differences between neostigmine and sugammadex use, we examined baseline characteristics of patients who received either neostigmine or sugammadex during common abdominal surgeries requiring intra- or perioperative NB

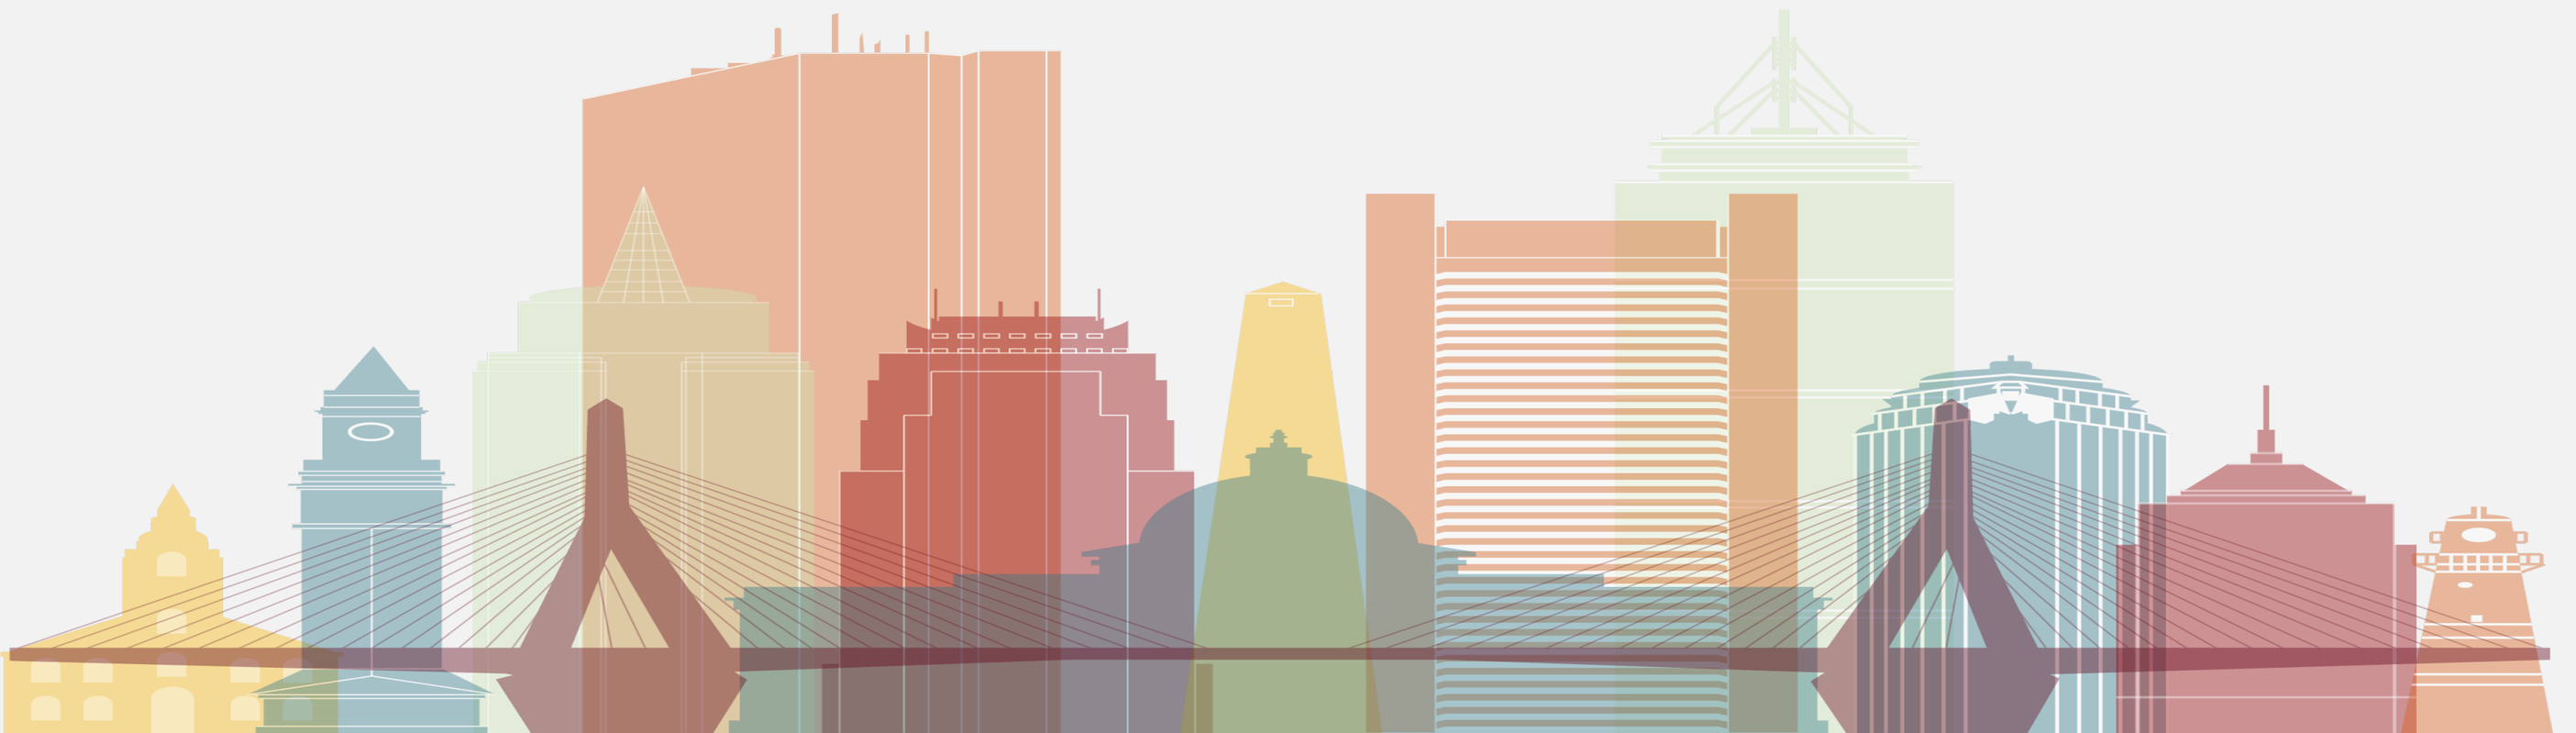

METHODS

- Using a large, de-identified US electronic health record database (Cerner Real-World Data), we extracted data for all adult patients (≥18 years) who underwent an inguinal hernia surgery, exploratory laparotomy, cholecystectomy, or appendectomy between January 2016 and May 2021, using ICD-10-PCS and Current Procedural Terminology codes
- We included those who received intra/perioperative rocuronium- or vecuronium-induced NB followed by sugammadex or neostigmine administration for NB reversal
- Sociodemographic factors, health insurance payor type, and Charlson Comorbidity Index (CCI) were compared across both sugammadex and neostigmine

RESULTS

- Of 74,088 patients who underwent target surgeries, 30,716 (41%) received sugammadex, including 41.8% of all males and 40.5% of all females
- Patients aged 60+ received sugammadex at higher rates than those aged 18-59 (43.4% vs. 40.3%), but neostigmine was more frequently administered than sugammadex across all age groups (**Figure 1**)
- Patients with BMI ≥30 received sugammadex at higher rates than those with BMI <18.5 (43.4% vs. 41.6%) (**Figure 2**)
- Patients with a CCI of 4+ received sugammadex more frequently than patients with scores of 0 (49.3% vs. 41.1%)
- A lower proportion of Hispanic/Latinx patients received sugammadex compared to all other ethnic groups (29.3% vs. 41.6%)
- A higher proportion of neostigmine recipients paid with commercial/private insurance compared to sugammadex recipients (57.4% vs. 54.1%), while a higher proportion of sugammadex recipients used Medicare/Medicaid compared to neostigmine recipients (33.7% vs. 31.7%) (**Figure 3**)

REFERENCES

1. Hristovska, A.-M., Duch, P., Allingstrup, M., & Afshari, A. (2017). Efficacy and safety of sugammadex versus neostigmine in reversing neuromuscular blockade in adults. *Cochrane Database of Systematic Reviews* , *8*, CD012763. https://doi.org/10.1002/14651858.CD012763. 2. Lee, Y. J., Money, K. L., & Elliott, A. (2019). Sugammadex compared with Neostigmine/Glycopyrrolate: An Analysis of Total PACU Time, Responsiveness, and Potential for Economic Impact. *INNOVATIONS in Pharmacy*, *10*(3), 1–1. https://doi.org/10.24926/iip.v10i3.1798. 3. Jiang, Y., Bash, L. D., & Saager, L. (2021). A Clinical and Budgetary Impact Analysis of Introducing Sugammadex for Routine Reversal of Neuromuscular Blockade in a Hypothetical Cohort in the US. *Advances in Therapy*, *38*(5), 2689–2708. https://doi.org/10.1007/s12325-021-01701-1. 4. Kheterpal, S., Vaughn, M. T., Dubovoy, T. Z., Shah, N. J., Bash, L. D., Colquhoun, D. A., Shanks, A. M., Mathis, M. R., Soto, R. G., Bardia, A., Bartels, K., McCormick, P. J., Schonberger, R. B., & Saager, L. (2020). Sugammadex versus Neostigmine for Reversal of Neuromuscular Blockade and Postoperative Pulmonary Complications (STRONGER): A Multicenter Matched Cohort Analysis. *Anesthesiology*, *132*(6), 1371–1381. https://doi.org/10.1097/ALN.0000000000003256. 5. Meghani, S. H., Byun, E., & Gallagher, R. M. (2012). Time to take stock: a meta-analysis and systematic review of analgesic treatment disparities for pain in the United States. *Pain Medicine* , *13*(2), 150–174. https://doi.org/10.1111/j.1526-4637.2011.01310.x. 6. Structural diagram of sugammadex sodium (https://upload.wikimedia.org/wikipedia/commons/c/c0/Sugammadex_sodium.svg) by Fvasconcellos, 9 January 2007. Public Domain.

** Analysis did not include patients for whom reimbursement data was unavailable*

FIGURE 1. Ratio of administration of sugammadex-to-neostigmine by age group

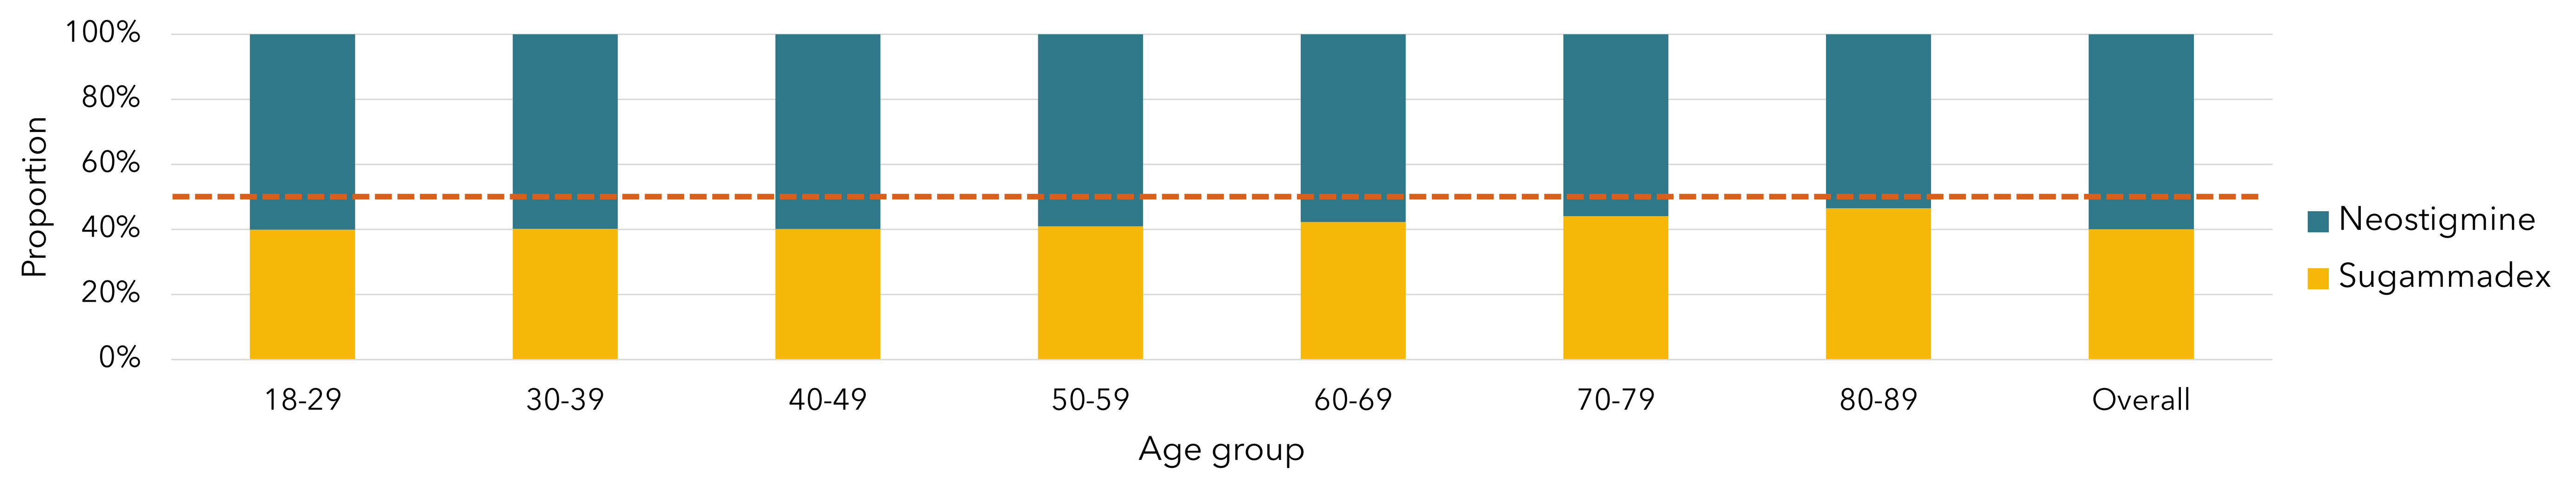
100%
80%
60%
40%
20%
0%
Proportion
18-29
30-39
40-49
50-59
60-69
70-79
80-89
Overall
Age group
Neostigmine
Sugammadex

CONCLUSIONS

- Age, ethnicity, BMI, CCI, and insurance payor type may influence the administration of sugammadex over neostigmine for NB reversal, potentially suggesting disparity in patient access
- Further research is required to understand whether other surgical procedures requiring NB reversal show similar trends in sugammadex use

FIGURE 2. Receipt of sugammadex vs. neostigmine by BMI

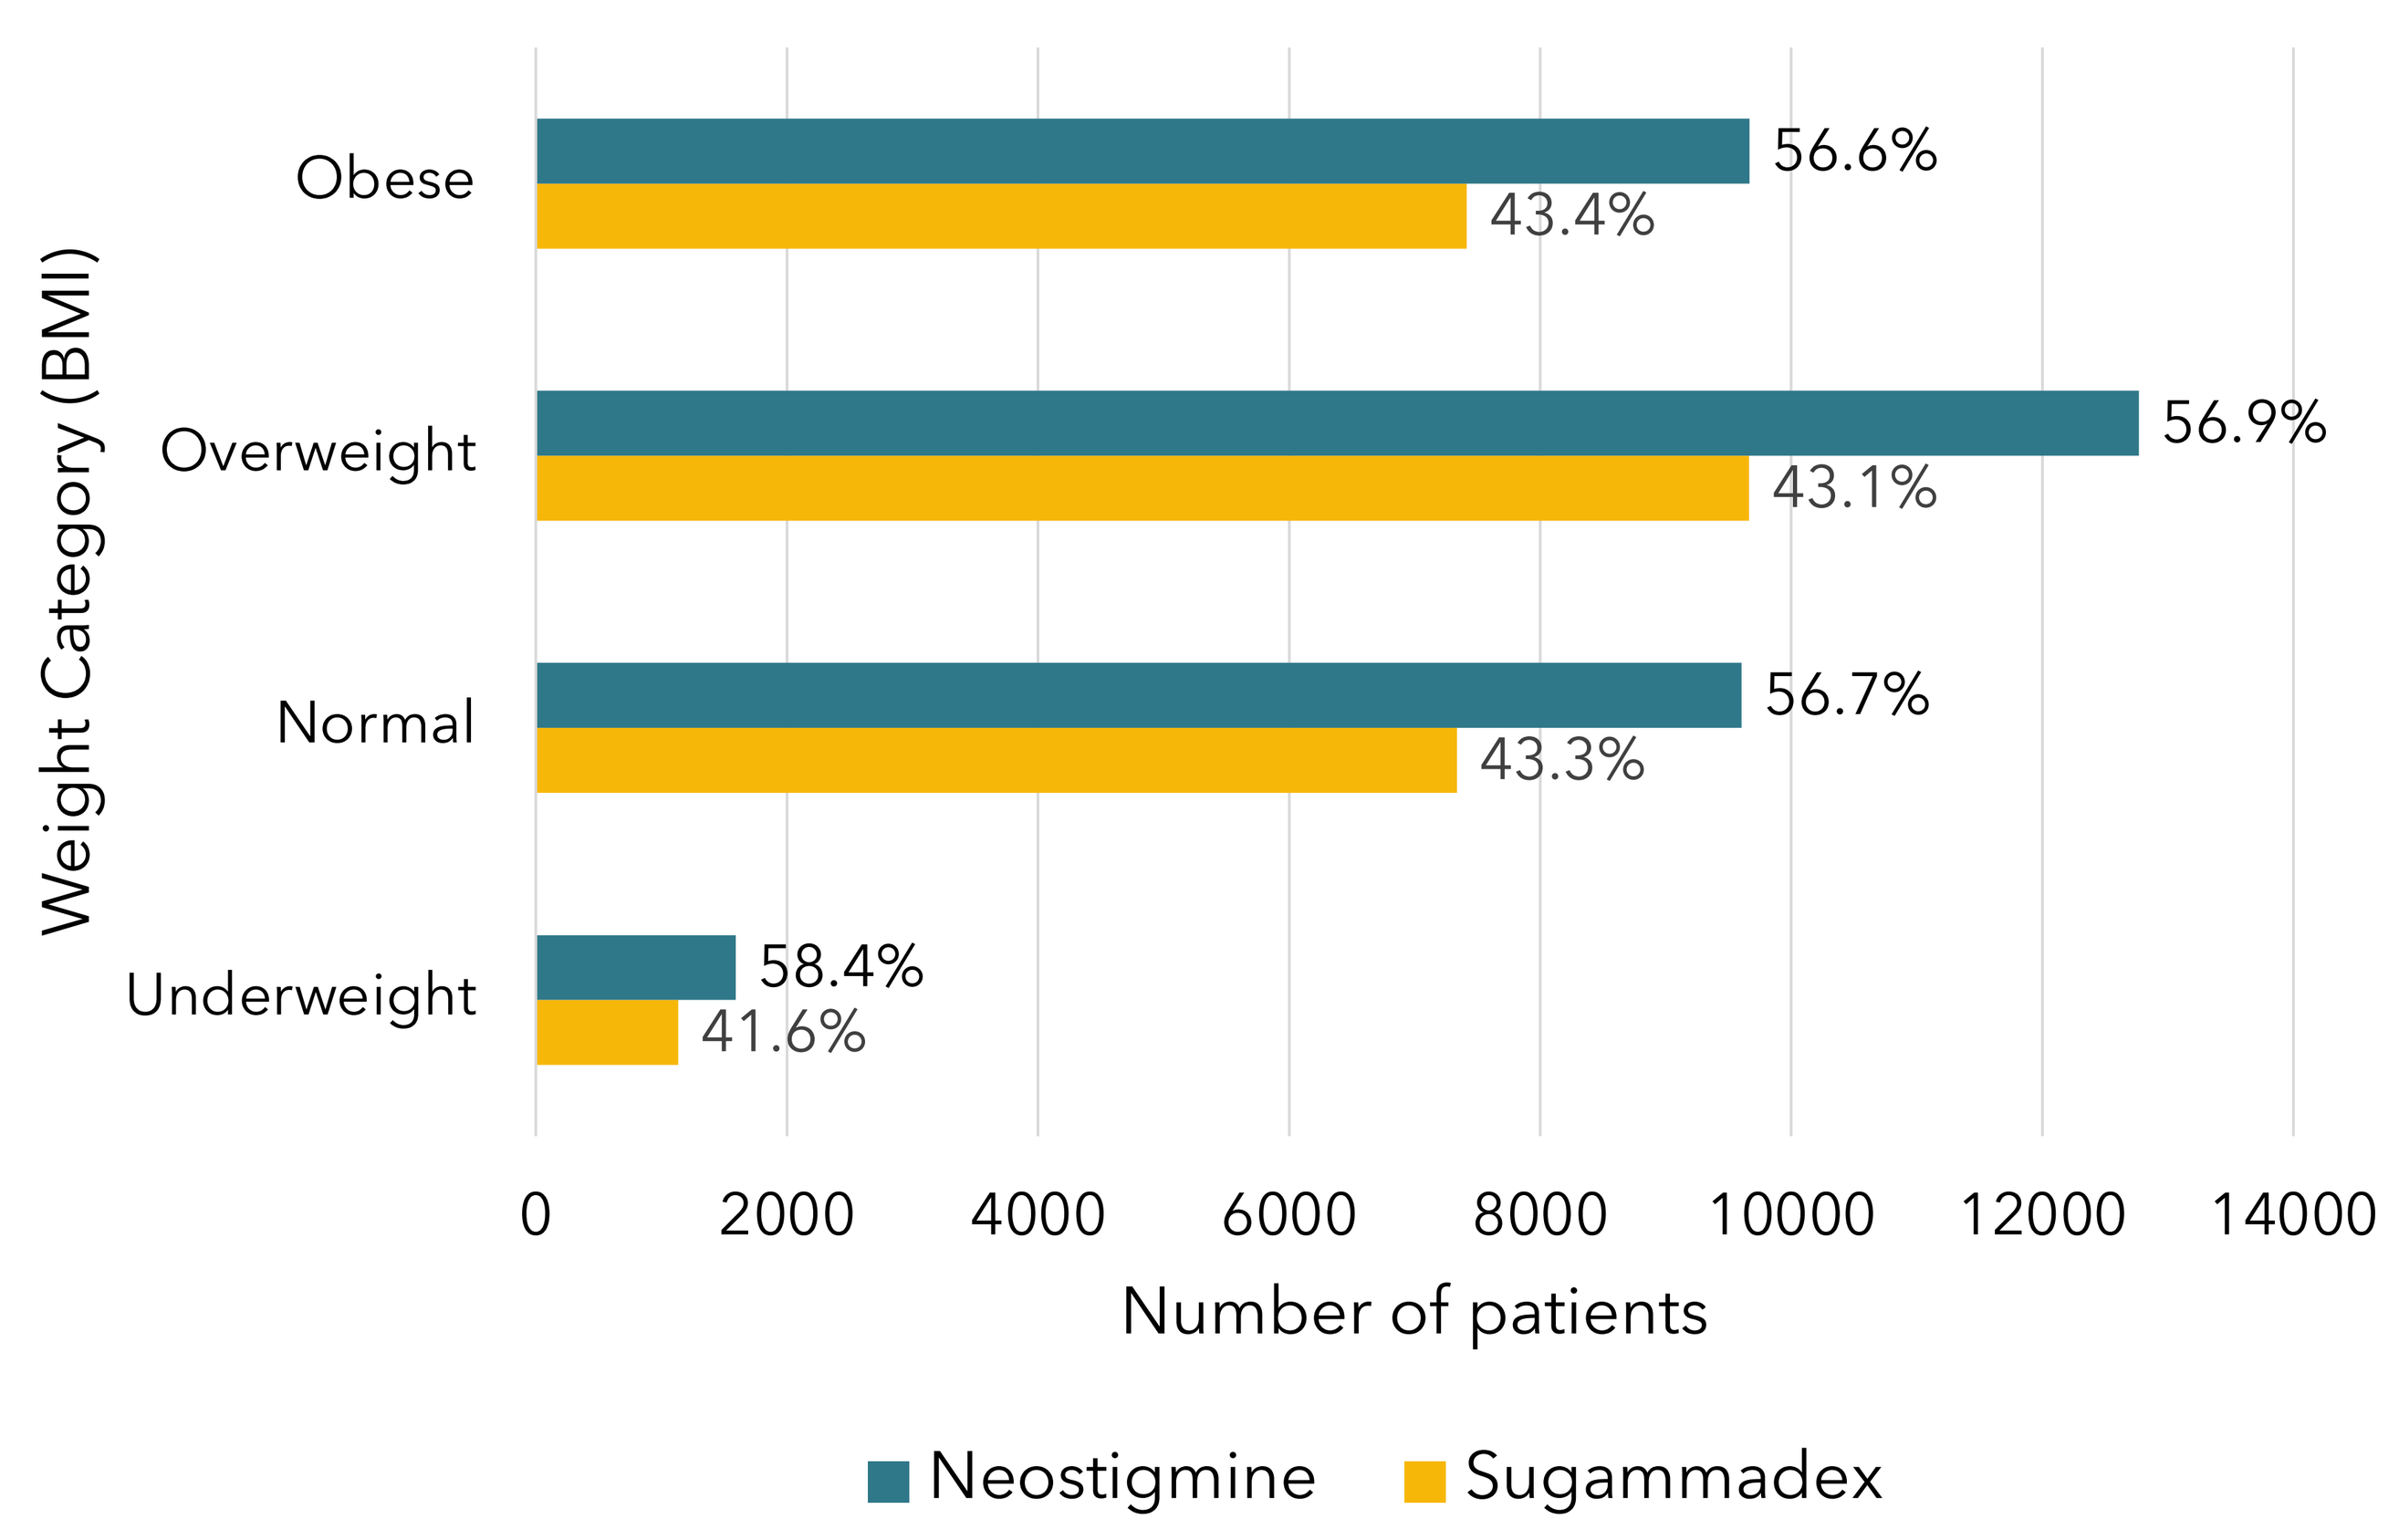
Obese
Overweight
Normal
Underweight
Weight Category (BMI)
56.6%
43.4%
56.9%
43.1%
56.7%
43.3%
58.4%
41.6%
0
2000
4000
6000
8000
10000
12000
14000
Number of patients
Neostigmine
Sugammadex

FIGURE 3. Sugammadex/neostigmine by health insurance payor type

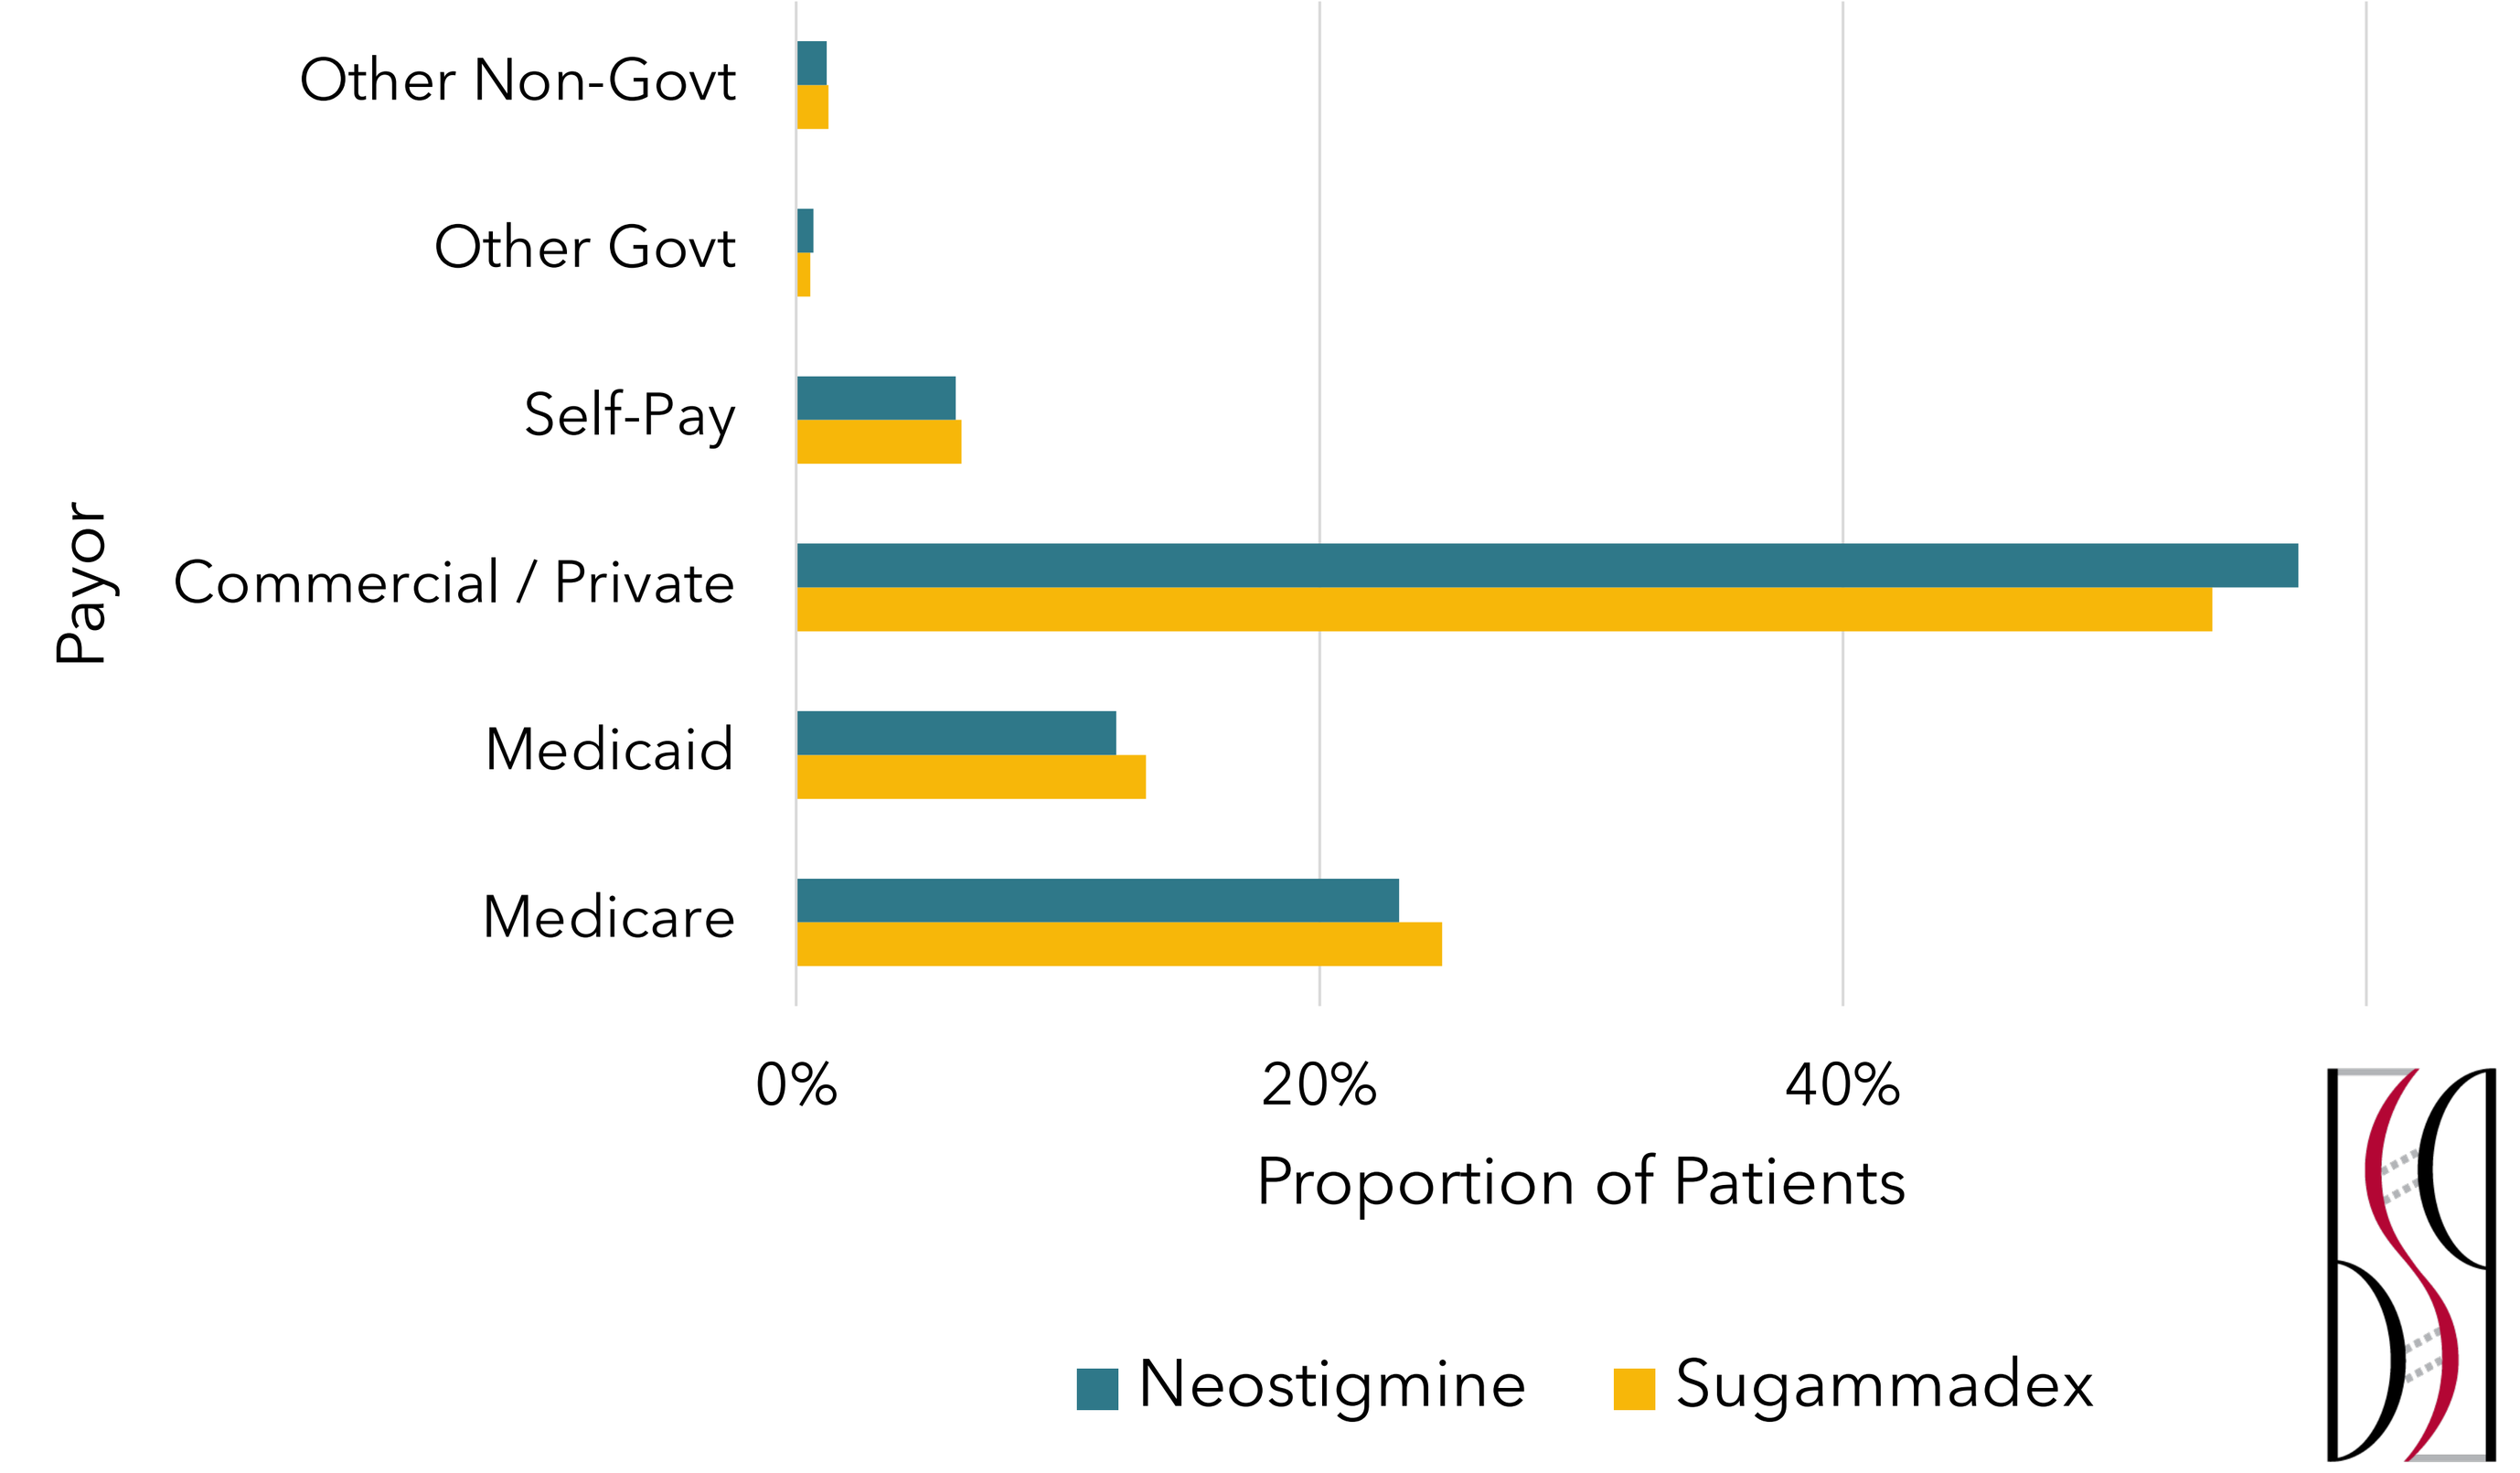
Other Non-Govt
Other Govt
Self-Pay
Commercial / Private
Medicaid
Medicare
Payor
0%
20%
40%
Proportion of Patients
Neostigmine
Sugammadex

HSD91