Early Real-World Experience with Emicizumab and Concomitant Factor VIII Replacement Products in Adult, Male People with Hemophilia A without Inhibitors

Lorraine Cafuir[1], Adina Estrin[2], Er Chen[3], David Hinds[3], Patricia Prince[2], Jennifer Thorburn[2], Henry Mead[3], Christine Kempton[1]

[1]Department of Hematology and Medical Oncology, Emory University School of Medicine, [2]Aetion, Inc., [3]BioMarin Pharmaceutical Inc.

Background

- Hemophilia A (HA) is a congenital X-linked bleeding disorder marked by a deficiency in clotting factor VIII (FVIII) affecting over 20,000, predominantly male, people in the United States (US)
- FVIII prophylaxis and recently approved emicizumab, a non-FVIII replacement therapy, are the standard of care for people with severe HA
- With the advent of novel therapeutics and limited studies on emicizumab, understanding its real-world use in adult, male people with HA (PwHA) without inhibitors is needed, including those switching from FVIII prophylaxis

Objective

- To describe patients who initiated emicizumab (new users), including cohort characteristics, use of concomitant FVIII, healthcare resource utilization (HCRU), and costs

Methods

- This study included adult, male PwHA without inhibitors who initiated emicizumab and were treated for at least 90 days and followed till censoring (mean follow-up of 12 months overall, 11.1 months for FVIII prophylaxis subgroup) **(Table 1)**
- Patients were required to be enrolled ≥ 180 days prior to and ≥ 90 days after index date
- First date of emicizumab use defined as index date

Table 1. Summary of study design

Dataset	IBM® Marketscan® (Dec 2015 – Dec 2020)
Population	Overall cohort: adults with HA without inhibitors Subgroup: those who were on prior FVIII prophylaxis (proxy for severe HA)
Index date	First emicizumab claim after Oct 4, 2018
Baseline	180 days prior to index date
Follow-Up	Variable follow-up, broken into induction (month 1) and maintenance periods (month 2+)
Censoring Reasons	Emicizumab treatment gap > 30 days, disenrollment, end of data (Dec 31, 2020)

Outcomes and Statistical Analysis

- Outcomes were measured using per-patient-per-month (PPPM) rates to account for variable follow-up time
 - HCRU: emicizumab and FVIII fills/in-office administrations and all-cause inpatient, outpatient, and ED visits
 - Costs inflated to 2020 US dollars using the medical component of the consumer price index

Sub-group Analysis- Prophylaxis Subgroup

- Patients meeting any of the following criteria at baseline were included in the FVIII prophylaxis subgroup:
 - ≥ 3 FVIII non-inpatient or non-emergency department (ED) fills/in-office administrations
 - Gaps in FVIII coverage of ≤ 60 days
 - ≥ 135 days of supply

Results

Patient Characteristics

- Among the 71 patients identified in the primary population **(Figure 1)** and the 52 patients in the FVIII prophylaxis subgroup, the mean age was 35 and 34 years, respectively
- Most patients were still actively taking emicizumab and enrolled at the end of the study period (62%)

Treatment and All-Cause HCRU

- 68% (FVIII prophylaxis subgroup: 73%) had at least one FVIII fill, with a mean of 0.37 (FVIII prophylaxis subgroup: 0.46) monthly fills during induction, and 0.17 (FVIII prophylaxis subgroup: 0.21) during maintenance **(Figure 2A)**
- There were, on average, 1.8 fills of emicizumab in the induction period and 1.0 monthly fills in the maintenance period **(Figure 2A)**
- Mean PPPM FVIII and emicizumab fills/administrations for the FVIII prophylaxis subgroup followed similar trends as the primary population, with slightly higher means across all time periods, as expected. The baseline period showed the largest difference (0.31) **(Figure 2A)**

Figure 1. Patient Attrition

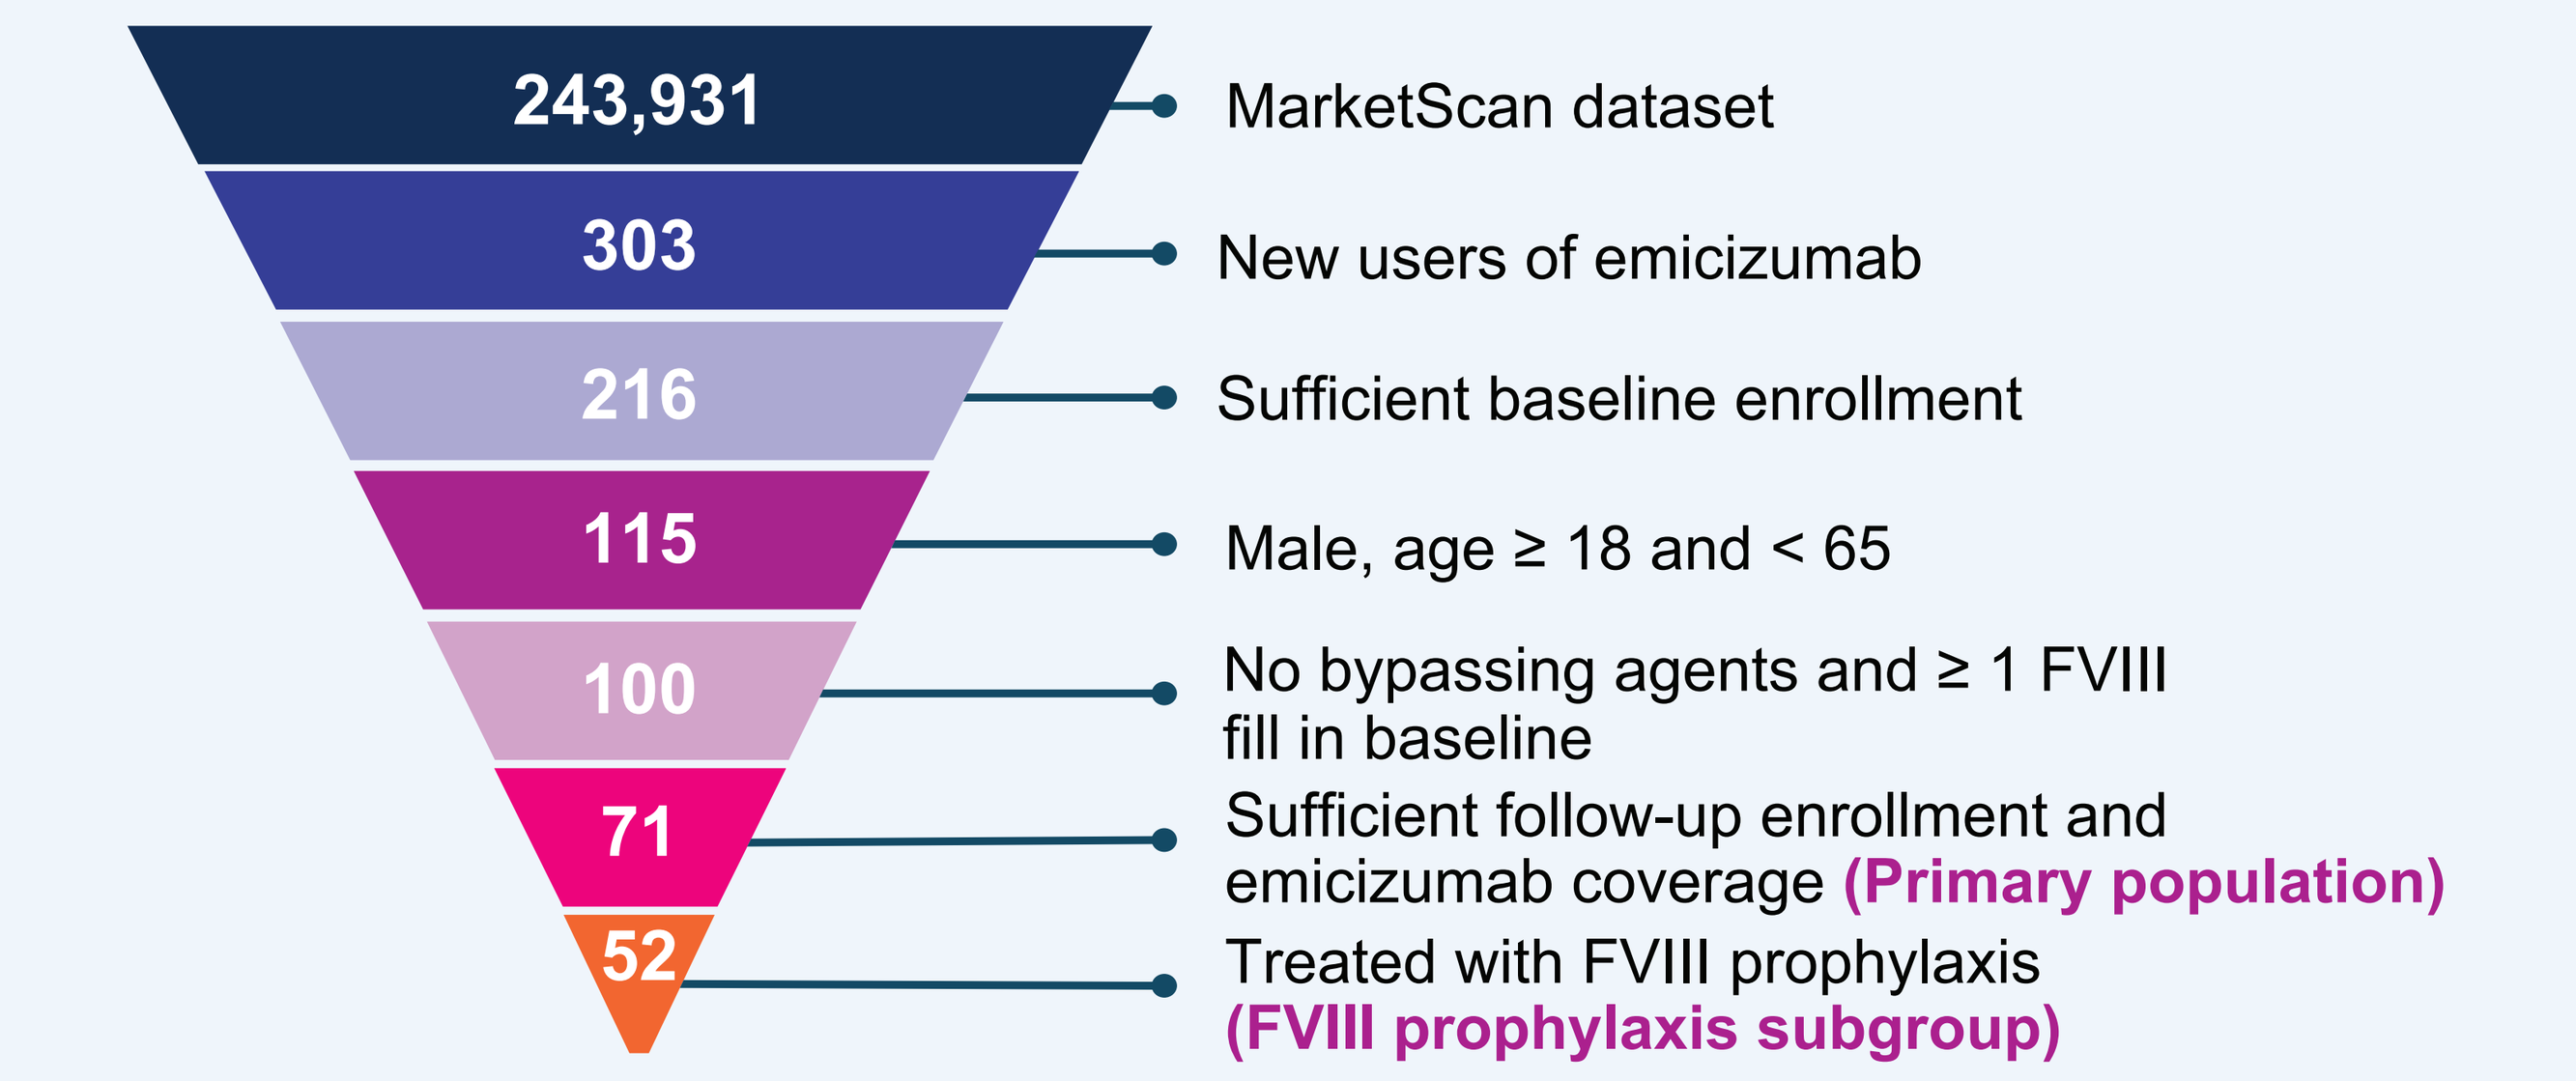
243,931
MarketScan dataset
303
New users of emicizumab
216
Sufficient baseline enrollment
115
Male, age ≥ 18 and < 65
100
No bypassing agents and ≥ 1 FVIII fill in baseline
71
Sufficient follow-up enrollment and emicizumab coverage (Primary population)
52
Treated with FVIII prophylaxis (FVIII prophylaxis subgroup)

Figure 2A. Mean PPPM FVIII and Emicizumab Fills/ Administrations

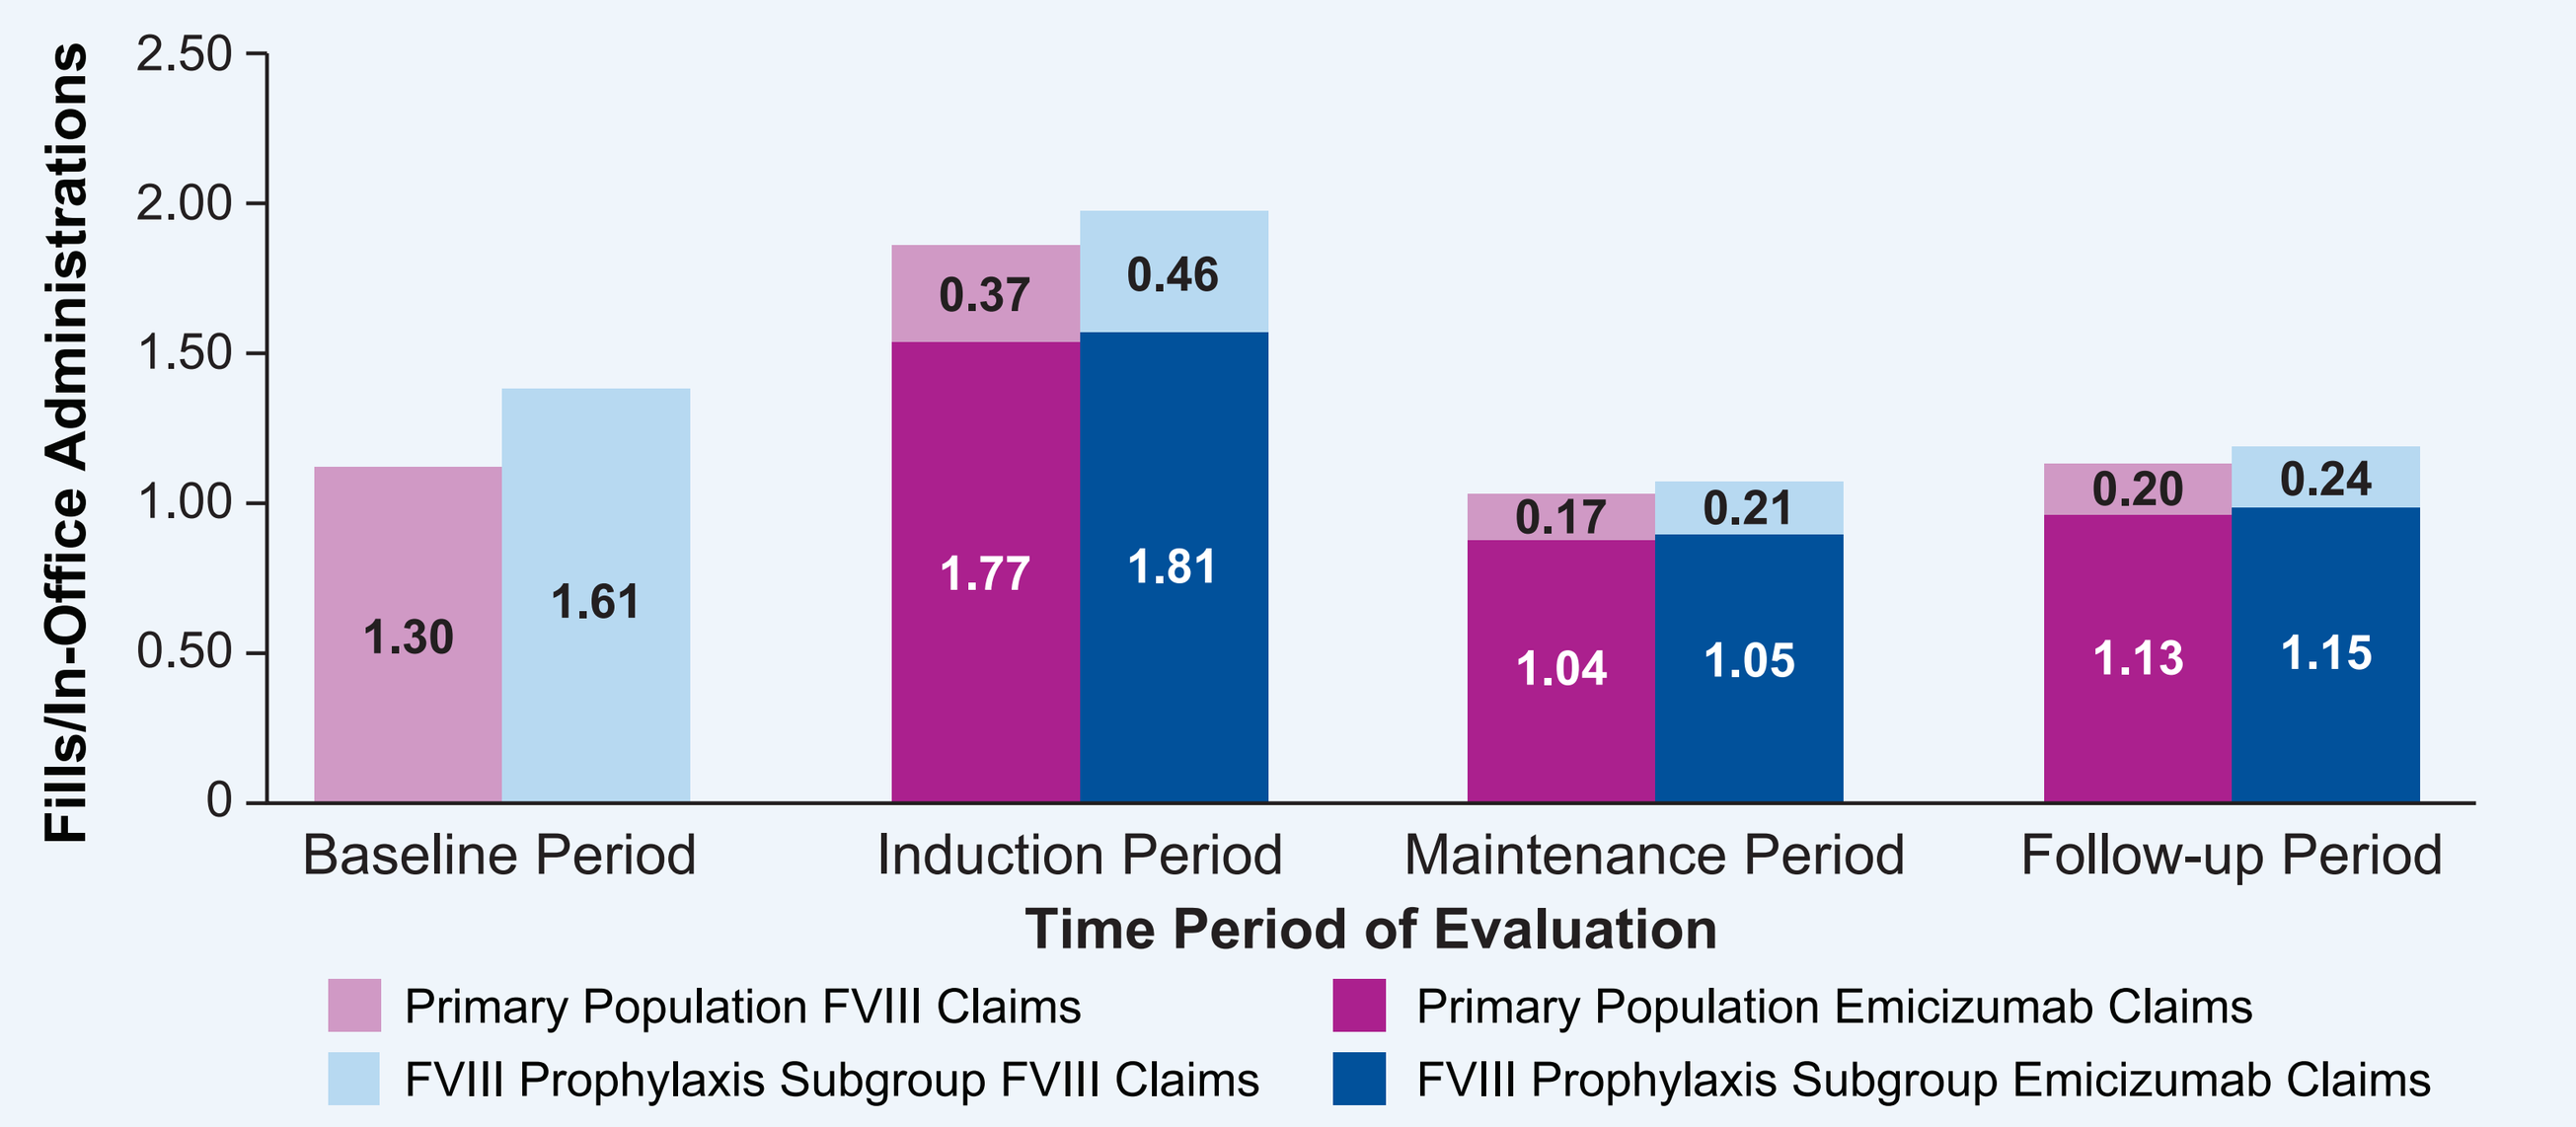
Fills/In-Office Administrations
2.50
2.00
1.50
1.00
0.50
0
1.30
1.61
0.37
0.46
1.77
1.81
0.17
0.21
1.04
1.05
0.20
0.24
1.13
1.15
Baseline Period
Induction Period
Maintenance Period
Follow-up Period
Time Period of Evaluation
Primary Population FVIII Claims
Primary Population Emicizumab Claims
FVIII Prophylaxis Subgroup FVIII Claims
FVIII Prophylaxis Subgroup Emicizumab Claims

Figure 2B. Mean PPPM Outpatient* Visits

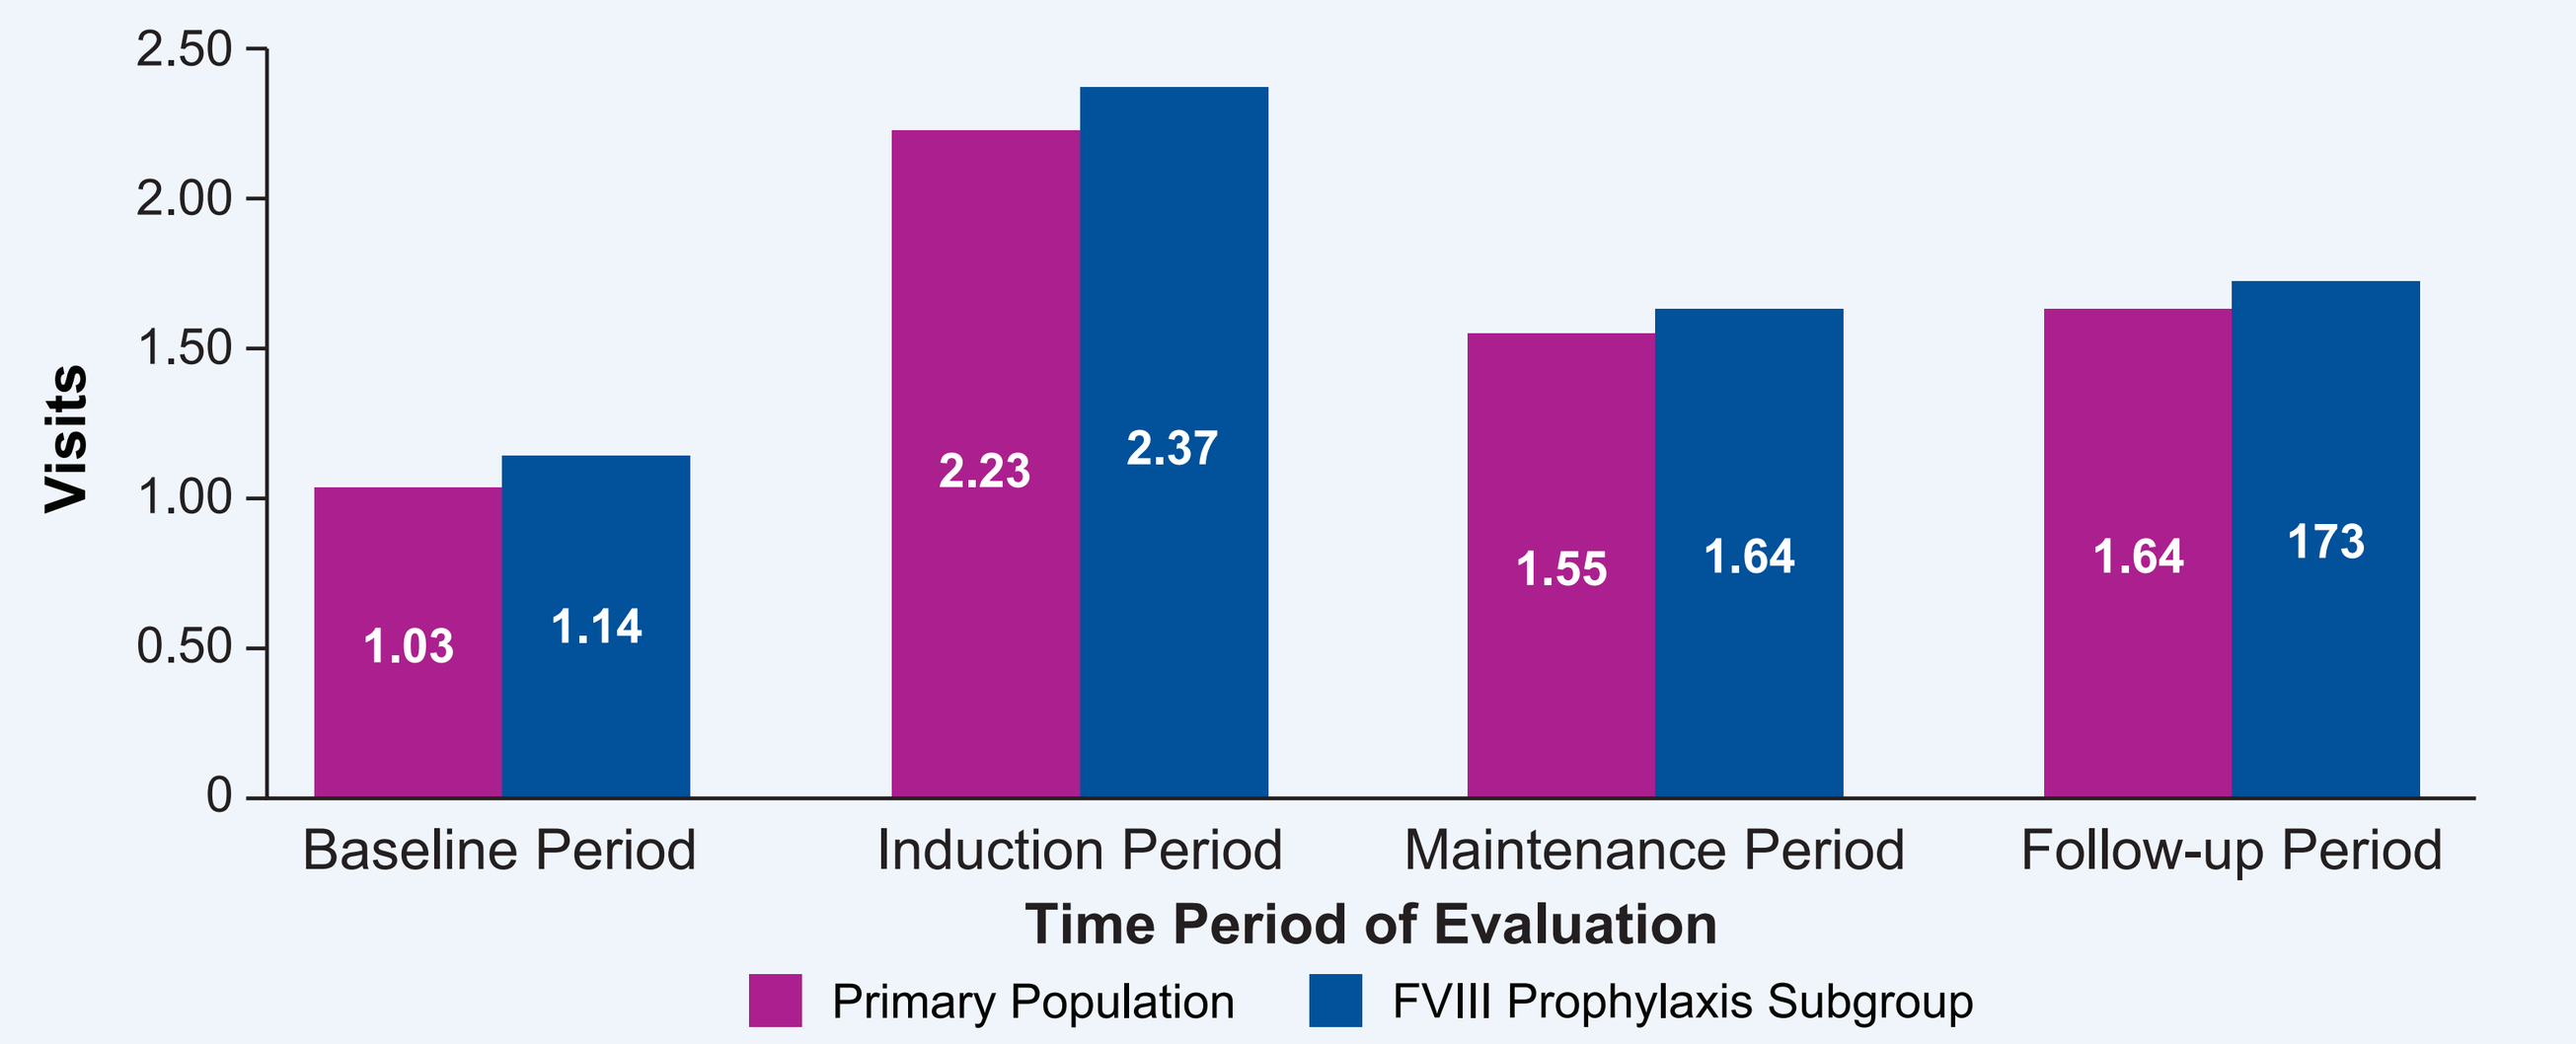
Visits
2.50
2.00
1.50
1.00
0.50
0
1.03
1.14
2.23
2.37
1.55
1.64
1.64
173
Baseline Period
Induction Period
Maintenance Period
Follow-up Period
Time Period of Evaluation
Primary Population
FVIII Prophylaxis Subgroup

*Non-FVIII-related outpatient visits

- Most patients had ≥1 non-FVIII-related outpatient encounter per month. Outpatient visits increased during emicizumab induction period (**Figure 2B**)
- 23% of patients had an ED visit; only one patient had an inpatient visit. None of these events were related to hemophilia or bleeding

Table 2. Mean Annualized Costs Over Baseline and Follow-Up

	Primary Population (FVIII Prophylaxis Subgroup)		
	Baseline Year	**First Year of Treatment***	**Subsequent Years of Treatment***
Emicizumab Costs	N/A	$726,681 ($731,657)	$642,623 ($644,438)
FVIII Costs	$506,710 ($622,535)	$50,491 ($61,512)	$40,444 ($47,556)
Total Healthcare Costs	$532,948 ($645,727)	$808,617 ($814,592)	$711,660 ($713,426)

*Projected costs; First year of treatment annualized costs = (Mean Induction Cost) + 11 *(Mean Maintenance Cost); Subsequent years of treatment annualized costs = (Mean Maintenance Cost) * 12

- 96% of total HCRU spent for the primary population (subgroup: 97%) was due to HA treatment **(Table 2)**
- Mean annualized total all-cause healthcare costs were $808,617 in the first year after emicizumab initiation for the primary population and $814,592 for the FVIII prophylaxis subgroup **(Table 2)**
- Mean annualized emicizumab costs in the first year were $726,681 for the primary population and $731,657 for the prior FVIII prophylaxis subgroup **(Table 2)**

Conclusions

- This study highlights the continued burden of treatment and healthcare costs for PwHA without inhibitors during the ~year following emicizumab initiation
- Patients in the FVIII prophylaxis subgroup, a proxy for severe HA, had higher FVIII use, HCRU, and costs than those in the primary population
- Key limitations include small sample sizes, administrative data, and limited follow-up duration
- This study cohort may not be representative of all PwHA taking emicizumab

Acknowledgments

BioMarin Pharmaceutical Inc. provided funding for the study, data analysis, writing, and poster production. All authors contributed to the development of the study protocol, analysis plan, and/or data interpretation, and to the preparation and review of the poster. All authors maintained control over the final content.

Presented at ISPOR annual conference: 15th–18th May 2022; National Harbor, MD (USA)

©2022 BioMarin Pharmaceutical Inc. All rights reserved.