Long-Term Outcomes of Lenacapavir plus Optimized Background Regimen versus Other Treatments for Multi-Drug Resistant HIV via a Cost-Effectiveness Model

Vardanega V[1], Rawlinson W[1], Francis-Graham S[1], Mezzio DJ[2], Eddowes LA[1]

[1]Costello Medical, London, United Kingdom, [2]Gilead Sciences, Foster City, California, United States

GILEAD

Background

- Achieving virologic suppression (VS) with combination antiretroviral (ARV) therapy is challenging for heavily treatment-experienced (HTE) people with HIV (PWH) due to multi-drug resistance (MDR), intolerance, or safety concerns.[1, 2]
- To maximize the probability of achieving VS, HTE individuals are often prescribed highly individualized treatment regimens, which require resistance testing as well as an assessment of treatment history, tolerability, past adherence and potential for drug-drug interactions.[3-5]
- An unmet need remains for new, effective and cost-effective treatment options for the management of PWH with MDR since these individualized regimens are often highly complex and not always effective.
- Lenacapavir (LEN), a first-in-class multi-stage HIV capsid inhibitor administered subcutaneously as a long-acting injectable (every 6 months), is under investigation in combination with an optimized background regimen (OBR) for treating PWH with MDR in the phase 2/3 CAPELLA study.[6,7]

Objective

To determine the long-term benefits (virologically suppressed life years [LYs], AIDS-free LYs and quality-adjusted life years [QALYs]) and associated costs resulting from the use of LEN+OBR vs fostemsavir (FTR)+OBR and ibalizumab (IBA)+OBR for the treatment of HTE PWH.

Table 1. Model overview and key model inputs

Model Overview	
Type of Model	Markov state-transition model
Country	US
Intervention	LEN+OBR
Comparators	IBA+OBR FTR+OBR
Population	PWH with MDR who are failing on their current therapy
Perspective	Third party healthcare payer
Time horizon (cycle length)	Lifetime (3 months)
Discount rate for costs and clinical outcomes	3%
Model Inputs	
Viral load parameters	CAPELLA[7] GS-US-183-0145[9] BRIGHTE[10] ITCs of LEN+OBR versus comparators[8]
CD4 cell count	CAPELLA[7] GS-US-183-0145[9] Mangal et al., 2017[11]
Drug acquisition and administration costs	LEN: WAC unavailable at the time of analyses (assumption) All other drugs: Red Book (WAC)[12] Administration costs: US Centers for Medicare and Medicaid Services Physician Fee Schedule[13]
Treatment switching costs	US Centers for Medicare and Medicaid Services Current Procedural Terminology, Fourth Edition[13]
OBR composition	Percentage of PWH receiving different components assumed equal to CAPELLA[7] OBR composition, removing IBA and FTR Dosing regimen: SmPCs[14] Pill size and cost: Red Book (WAC)[12]

Abbreviations: FTR, fostemsavir; IBA, ibalizumab; ITC, indirect treatment comparison; LEN, lenacapavir; MDR, multidrug resistance; OBR, optimized background regimen; PWH, people with HIV; SmPC, summary of product characteristics; US, United States; WAC, wholesale acquisition cost.

Methods

Model Framework

- A Markov state-transition model with a US healthcare payer perspective and lifetime time horizon was developed to evaluate the costs and benefits of LEN+OBR vs FTR+OBR and IBA+OBR in PWH with MDR. An overview of the model is provided in **Table 1**.
- PWH occupied health states stratified by viral load and CD4 cell count categories within two lines of therapy: intervention/comparator and salvage therapy **(Figure 1)**.
- Rate of change in viral load determined transition to salvage therapy, which could be delayed by effective treatments.
- Viral load influenced CD4 cell count, which in turn determined mortality rates, health state utilities and healthcare resource use costs.

Model Inputs

- Transition probabilities between viral load categories were based on extrapolated viral load data (up to Week 52) from the CAPELLA study for LEN+OBR.[7]
- For comparators, transition probabilities were based on relative efficacy parameters obtained from indirect treatment comparisons (ITCs).[8]
- CD4 cell count transition probabilities were dependent on viral load (PWH with viral load >200 copies/mL are more likely to transition to lower CD4 cell counts than virologically suppressed PWH) and assumed to remain constant over time.
- Additional model inputs were informed by Week 52 data from the CAPELLA trial,[6,7] systematic and targeted literature reviews, discussion with clinical experts and internal sources. Key inputs are summarized in **Table 1** along with their respective sources.

Model Outputs

- The key measures of health benefits included undiscounted total virologically suppressed LYs, AIDS-free LYs and QALYs. VS was defined as <50 copies/mL and AIDS as CD4 cell count <200 cells/mm^3.
- Sensitivity analyses were performed to address elements of uncertainty in the model and to explore the robustness of the results.
- The principal drivers of incremental QALYs for each comparison were identified using deterministic sensitivity analyses (DSA).
- A threshold analysis was also performed to determine cost-effectiveness at different willingness-to-pay (WTP) thresholds.

Results

- LEN+OBR was estimated to extend the number of LYs spent virologically suppressed, increase LYs spent AIDS-free and lead to the accrual of more QALYs in PWH with MDR, vs the comparator regimens. The estimated health benefits (undiscounted) in terms of virologically suppressed LYs, AIDS-free LYs and QALYs gained are presented in **Figure 2**.
- In the base case model, LEN+OBR was associated with lower medical costs due to treatment switching and AIDS related events, over the patient's lifetime versus all comparators. Higher disease management costs were observed for LEN+OBR due to increased LYs and staying on HIV treatment longer before failing and switching to salvage therapy.
- DSA demonstrated that the key inputs driving the incremental QALYs for LEN+OBR vs FTR+OBR were LEN+OBR risk ratios for viral load >200 copies/mL and the extrapolation parameters used to derive LEN+OBR viral load **(Figure 3)**. DSA for LEN+OBR vs IBA+OBR showed similar trends.

Figure 2. Health benefits (undiscounted) for LEN+OBR vs comparators

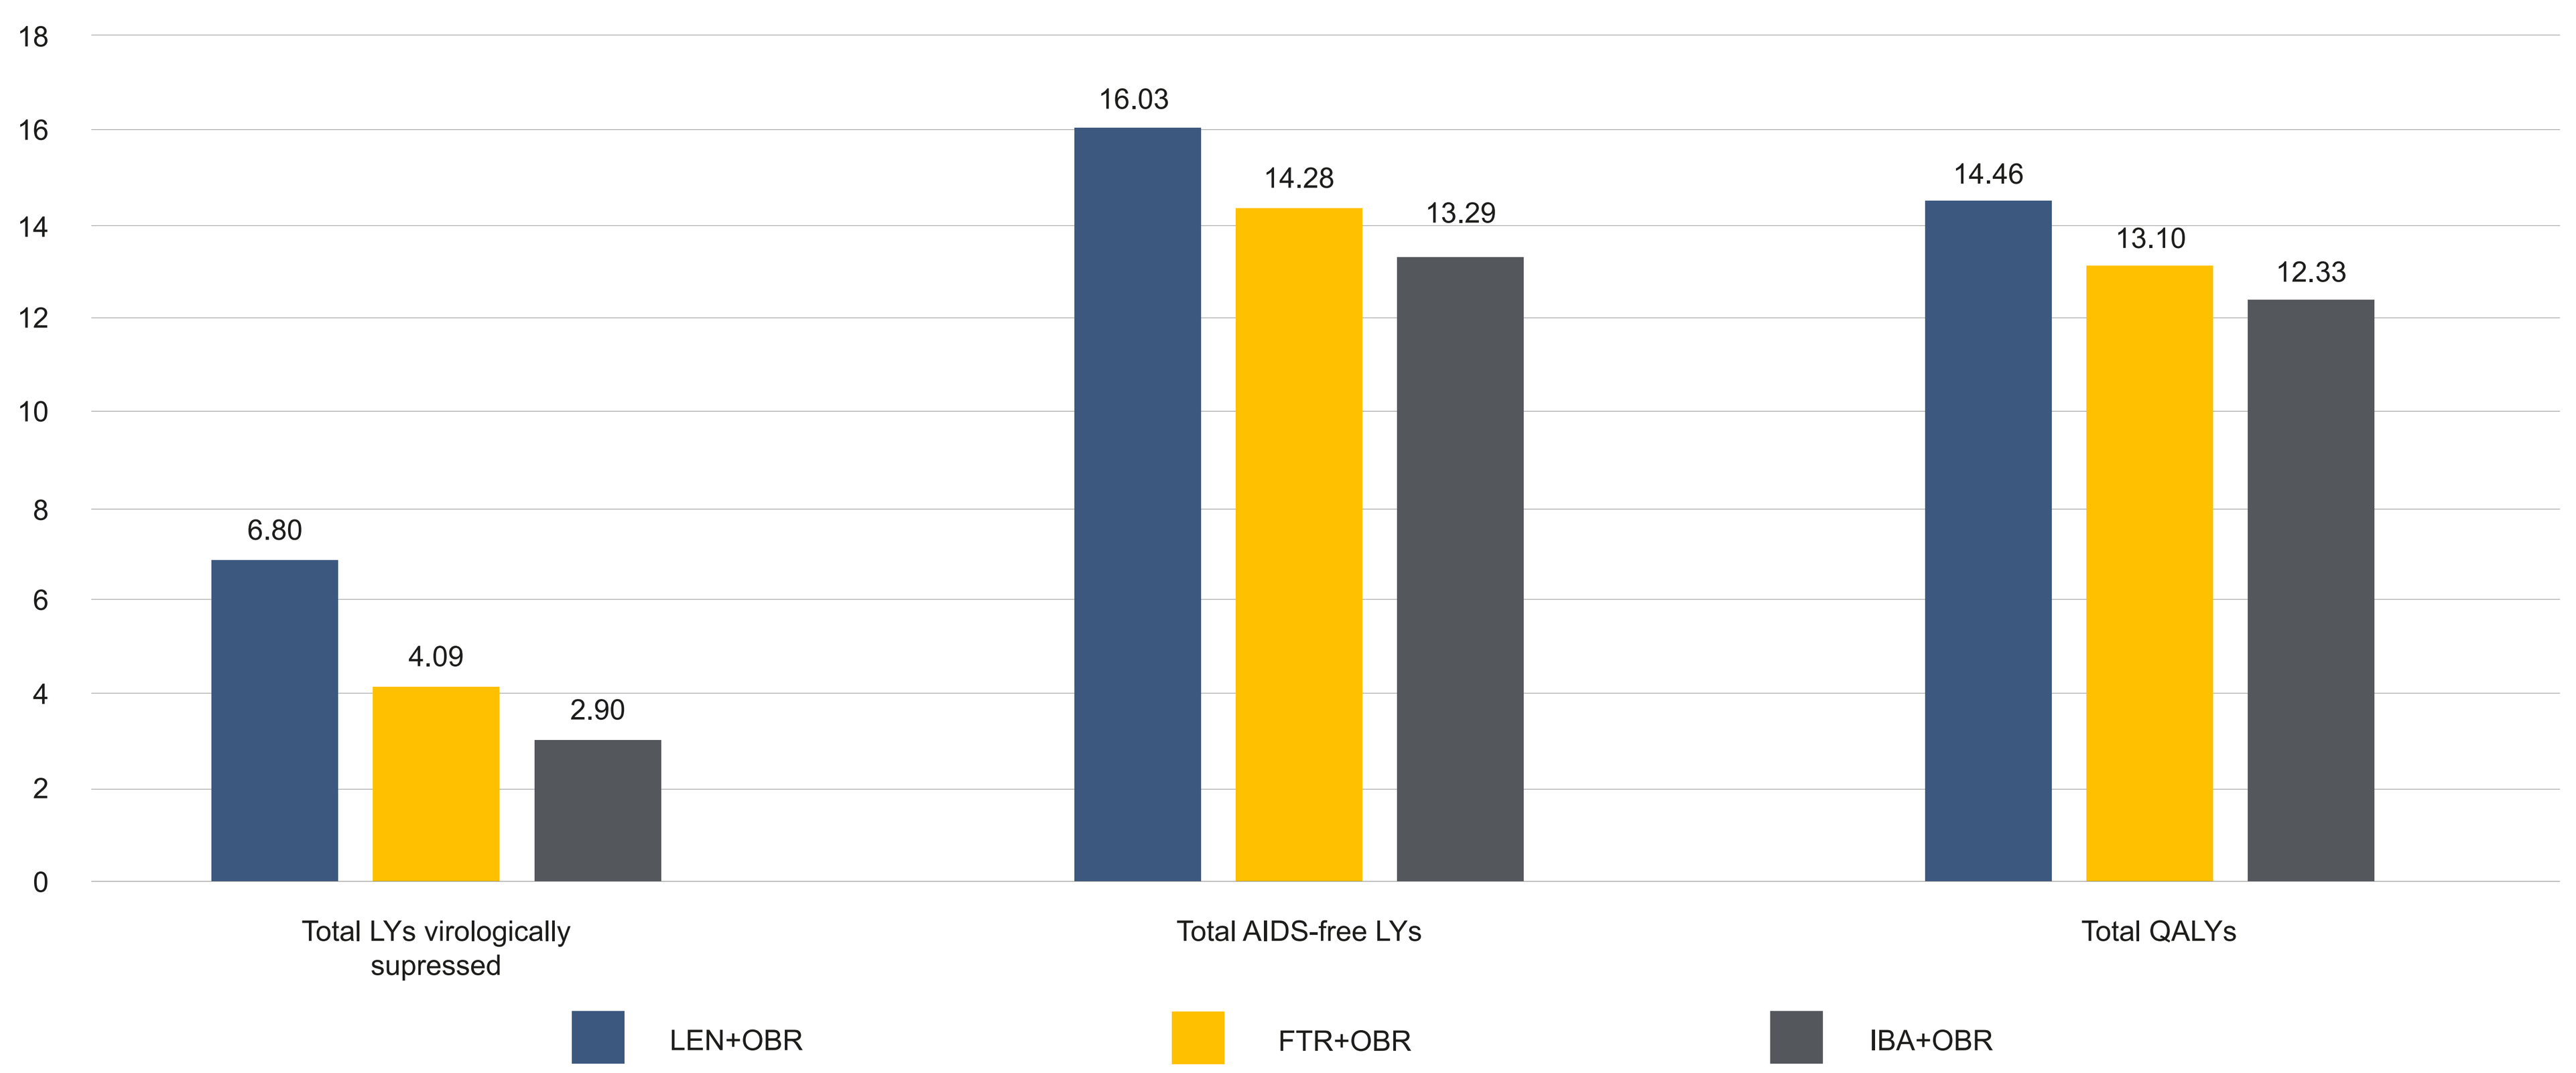
18
16
14
12
10
8
6
4
2
0
6.80
4.09
2.90
16.03
14.28
13.29
14.46
13.10
12.33
Total LYs virologically supressed
Total AIDS-free LYs
Total QALYs
LEN+OBR
FTR+OBR
IBA+OBR

Abbreviations: AIDS, acquired immunodeficiency syndrome; FTR, fostemsavir; IBA, ibalizumab; LEN, lenacapavir; LYs, life years; OBR, optimized background regimen; QALYs, quality-adjusted life years.

Figure 1. Overall model structure and health states within each therapy line

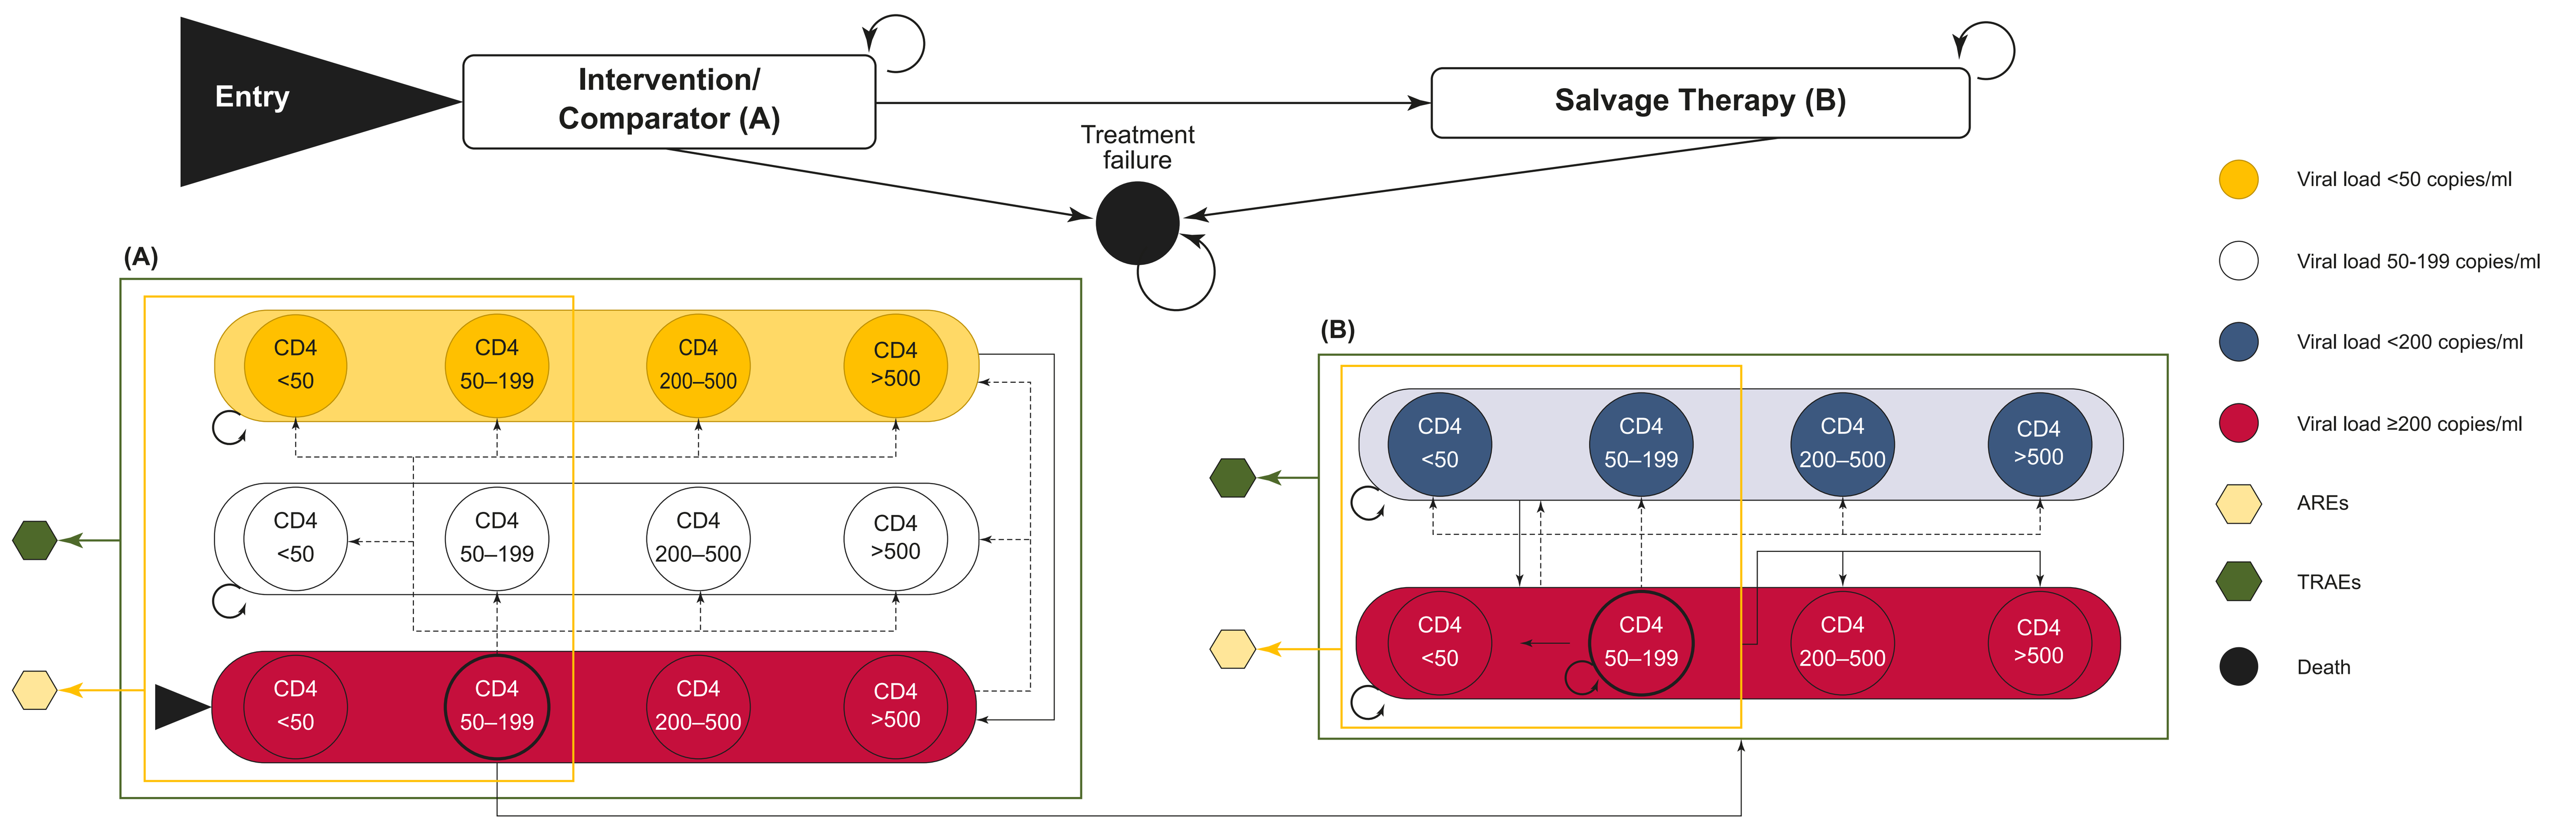
Entry
Intervention/ Comparator (A)
Salvage Therapy (B)
Treatment failure
(A)
(B)
CD4 <50
CD4 50–199
CD4 200–500
CD4 >500
Viral load <50 copies/ml
Viral load 50-199 copies/ml
Viral load <200 copies/ml
Viral load ≥200 copies/ml
AREs
TRAEs
Death

Abbreviations: AIDS, acquired immunodeficiency syndrome; AREs, AIDs-related events; CD4, cluster of differentiation 4; TRAEs, treatment-related adverse events. **Footnotes:** Solid arrows represent allowed transitions across viral load categories and lines of therapy; dashed arrows show transitions that can occur only in the first cycle after initiation of the treatment line; circular arrows indicate that PWH can remain in the state for more than one cycle. Viral load categories are further stratified into CD4 cell count categories. CD4 cell count-related health states are color-coded to indicate the respective viral load category. Health states are represented with a circle shape, events with a hexagon shape. For the intervention/comparator line, transitions allowed from only one health state i.e. the CD4 cell count 50-199/viral load ≥200 copies/mL (A) are shown as an example for how PWH could flow through the model for the intervention/comparator and salvage therapy lines; similar transitions from other health states were also possible. Those in the intervention/comparator line were not able to transition from viral load 50–199 copies/mL back to <50 copies/mL in the model, due to limited data on this transition. PWH could transition out the intervention/comparator line and move into salvage therapy. For the salvage therapy line, only transitions allowed from one health state are shown i.e. the CD4 cell count 50-199/viral load ≥200 copies/mL health state (B). Using default model settings, no PWH with MDR could achieve virologic suppression (e.g. viral load <200 copies/mL) in the salvage therapy line. PWH who commenced salvage therapy were assumed to remain there until death in the model. AREs and TRAEs were events that PWH could experience without leaving their current health state. PWH can transition to the self-absorbing death state from all health states; mortality is adjusted depending on age and CD4 cell count.

Figure 3. Tornado plot showing results of the incremental QALYs DSA (LEN+OBR vs FTR+OBR)

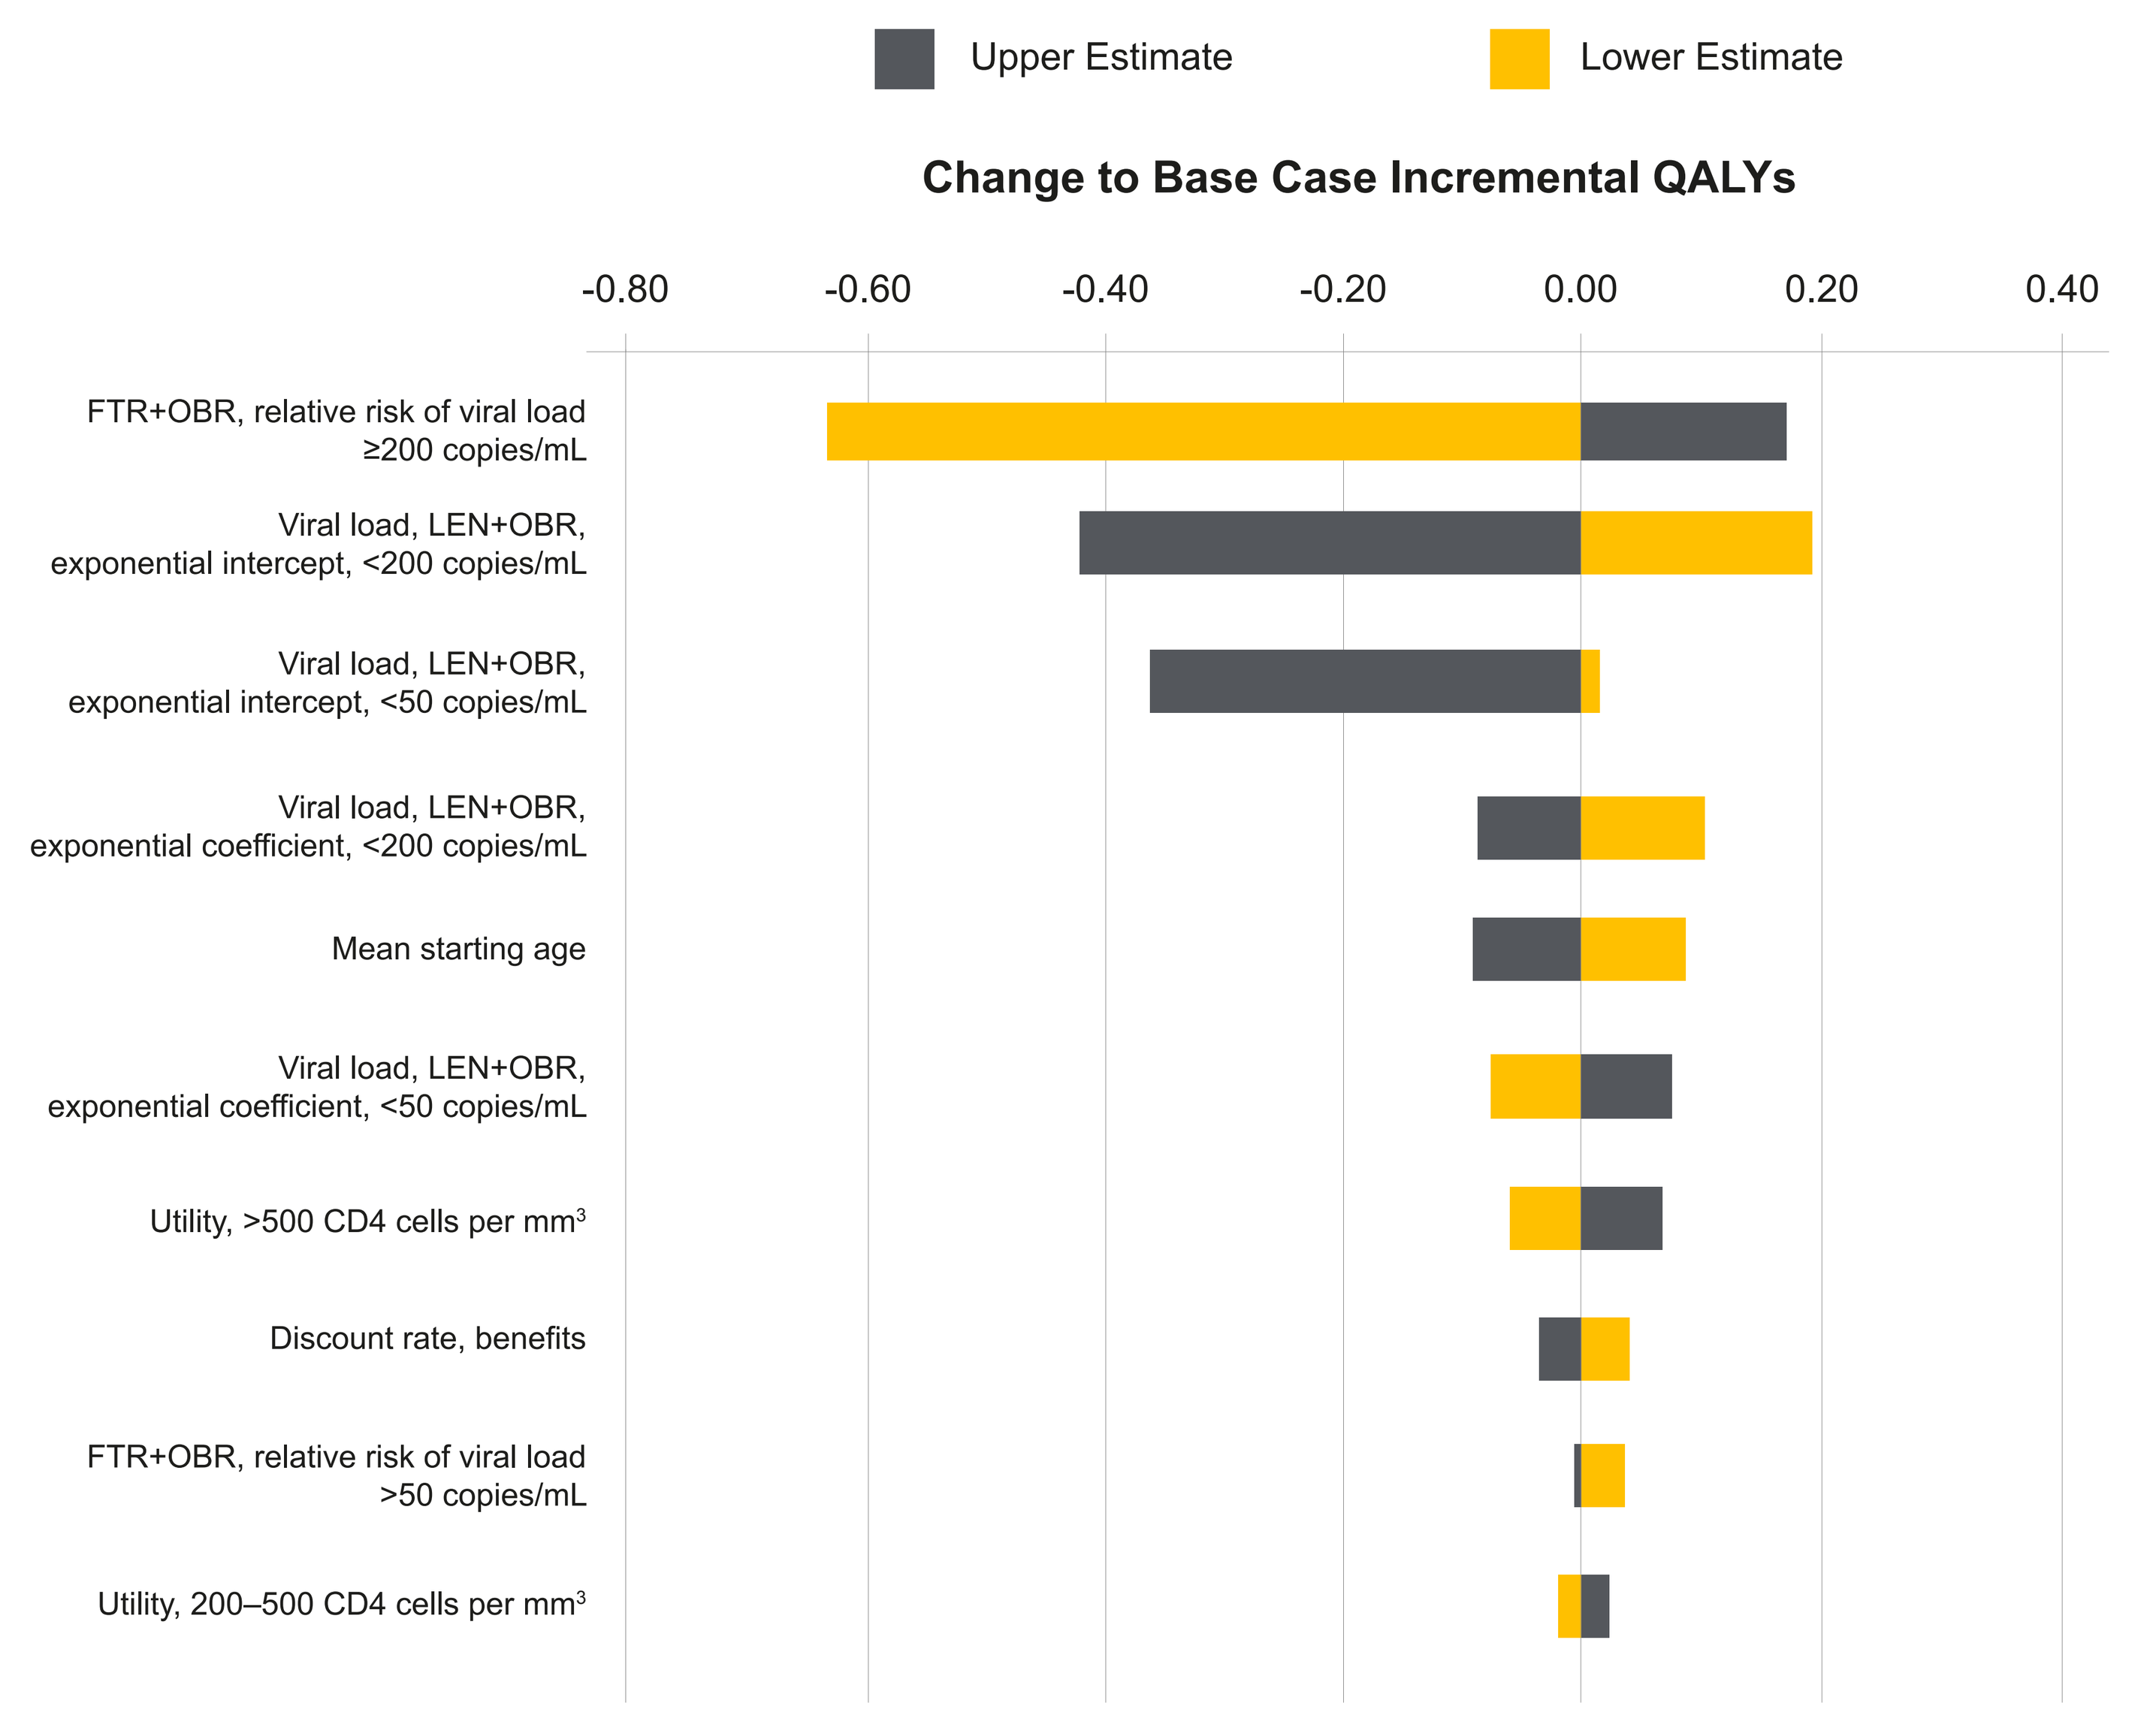
Upper Estimate
Lower Estimate
Change to Base Case Incremental QALYs
-0.80
-0.60
-0.40
-0.20
0.00
0.20
0.40
FTR+OBR, relative risk of viral load ≥200 copies/mL
Viral load, LEN+OBR, exponential intercept, <200 copies/mL
Viral load, LEN+OBR, exponential intercept, <50 copies/mL
Viral load, LEN+OBR, exponential coefficient, <200 copies/mL
Mean starting age
Viral load, LEN+OBR, exponential coefficient, <50 copies/mL
Utility, >500 CD4 cells per mm3
Discount rate, benefits
FTR+OBR, relative risk of viral load >50 copies/mL
Utility, 200–500 CD4 cells per mm3

Abbreviations: FTR, fostemsavir; LEN, lenacapavir; OBR, optimized background regimen; QALYs, quality-adjusted life years.

Limitations and Challenges

- Clinical data for PWH with MDR are limited, partly due to the small population size and heterogeneity within this population. Therefore, there are some uncertainties related to the derived probabilities for transitions between different health states.
- Not all possible OBR compositions, treatment switches, costs associated with switching treatment components and the effect of these variables on treatment efficacy could be captured in the model. However, the inputs were carefully chosen based on the available literature and clinical expert opinion, to simulate conditions that closely reflect the clinical reality of treating PWH with MDR, and these inputs can be easily adapted to reflect the changing HIV treatment landscape.

Conclusions

The model results indicate that, compared to its key comparators, LEN+OBR is associated with improved long-term clinical benefits in terms of virologically suppressed LYs, AIDS-free LYs and QALYs gained, and has the potential to delay transition to salvage therapy in HTE PWH.

References: **1.** Zdanowicz M. A Am J Pharm Educ 2006;70; **2.** Cutrell J. *et al.* Ther Adv Inf Dis 2020;7; **3.** British HIV Association (BHIVA). BHIVA guidelines for the treatment of HIV-1-positive adults with antiretroviral therapy 2016; **4.** European AIDS Clinical Society (EACS). Guidelines Version 10.1 October 2020; **5.** Saag MS. *et al.* Jama 2020;324:1651-1669; **6.** Segal-Maurer S. *et al.* CROI 2021; Abstract 127; **7.** Gilead Data on File. CAPELLA Week 52 CSR 2022; **8.** Chatzidaki I. *et al.* AMCP 2022; **9.** Gilead Data on File 2012; **10.** Kozal M. *et al.* New Engl J Med 2020;382:1232-1243; **11.** Mangal TD. *et al.* AIDS 2017;31:1073-1082; **12.** IBM. Micromedex Red Book Feb 2022; **13.** US Centers for Medicare & Medicaid Services. Physician fee schedule 2021; **14.** Gilead Data on File. EMA.Lenacapavir Draft SmPC.

Author Contributions: All authors contributed to the study conception/design, acquisition/analysis/interpretation of data, drafting/critically revising for important intellectual content and the final approval of the publication.

Acknowledgements: This study was funded by Gilead Sciences. The authors acknowledge Vaishali Yadav, PhD (Costello Medical, UK) for medical writing and editorial assistance. All costs associated with development of this poster were funded by Gilead Sciences.

To be presented at ISPOR 2022 | Washington, DC, USA | May 15–18, 2022

© Gilead Sciences, Inc. All rights reserved