EE301

Improvement in Work Productivity Loss and Associated Costs for Crohn's Disease Patients Receiving Ustekinumab in the United States: Findings From a Real-World Survey

Z. Ding[1], E. Muser[1], J. Izanec[1], A. Roughley[2], R. Lukanova[2], J. Kershaw[2]

[1]Janssen Scientific Affairs, LLC., Horsham, PA, USA; [2]Adelphi Real World, Bollington, UK

Scan the QR code

https://www.congresshub.com/Immunology/ISPOR2022/Ustekinumab/Ding

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

BACKGROUND

- Work-productivity loss (WPL) is an important contributor to the economic burden of Crohn's disease (CD).
- The Short Inflammatory Bowel Disease Questionnaire (SIBDQ) is a validated questionnaire used to assess Health Related Quality of Life (HRQoL), commonly used amongst CD patients.
- The EuroQol 5 Dimension Questionnaire (EQ-5D) is a non-disease-specific tool, widely used to measure HRQoL of patients.
- Ustekinumab (UST) is a biologic approved for treating adults with moderate-to-severe CD.

OBJECTIVE

- We aimed to report WPL and associated costs in patients with CD receiving UST, relative to those not receiving UST, in the real world.

METHODS

- Data were collected using the Adelphi Inflammatory Bowel Disease-Disease Specific Programme™ [1], a point in time study of gastroenterologists and their CD patients in the United States.
- Data were collected from 3 waves. The 2015 and 2017 waves (merged) captured CD patients who were not receiving UST and had moderate-to-severe disease state at the time of consultation (defined by Crohn's Disease Activity Index (CDAI) >220). The 2021 wave captured CD patients receiving UST at the time of consultation.
- Gastroenterologists completed patient record forms for their next 5 – 10 consulting CD patients, recording age, disease duration, employment status, and clinical status (CDAI score).
- Patients completed a voluntary self-completion (PSC) form capturing patient reported outcomes including the Short Inflammatory Bowel Disease Questionnaire (SIBDQ) (for each question, score range: 1 = poor HRQoL, 7 = optimum HRQoL)[2], EuroQoL 5-dimension questionnaire (EQ-5D) (score range: 0 = death, 1 = perfect health)[3], and Work Productivity Activity Impairment (WPAI)[4] questionnaire.
- All patients included in this study had completed a WPAI questionnaire.
- Mean WPL was calculated using the WPAI measures, annual (USD) lost from WPL per patient was calculated using the equations below. It was calculated per patient and then the mean was taken. A mean salary of $57,900[5] and 2080 working hours in a year were assumed.
 - Total WPL-related costs = absenteeism costs + presenteeism costs
 - Absenteeism costs = $57,900*work time missed
 - Presenteeism costs = $57,900*(1 – work time missed)*(productivity impairment)

RESULTS

- 39 CD patients receiving UST and 39 CD patients not receiving UST were included. Mean age was 41.1 and 39.1 with mean disease duration of 3.9 and 5.6 years for the UST and non-UST patient groups, respectively. 38.5% of UST patients had not received a biologic before.

Table 1. Physician-Reported UST Patient Characteristics at the Time of Consultation

	Patients receiving UST (n=39)	Patients not receiving UST (n=39)
Mean age [median] (SD)	41.1 [41.0] (10.9)	39.1 [39.0] (12.1)
Patient sex, n (% male)	21 (53.8)	21 (53.8)
Disease duration (years) [median] (SD)	3.7 [2.7] (3.9)	5.6 [3.0] (6.2)
BMI [median] (±SD)	25.0 [25.5] (2.8)	24.9 [24.7] (4.6)
Patient-reported satisfaction with control of their condition, n (% satisfied)	35 (89.7)	16 (41.0)
Current treatment*, n (%)		
Corticosteroid	10 (25.6)	11 (28.2)
Immunomodulator	6 (15.4)	17 (43.6)
Biologic	39 (100.0)	23 (60.3)

*Patients can be receiving more than 1 treatment, so %'s may add over 100%

- 91.0% of UST and 69.0% of non-UST patients were not currently experiencing a flare (**Figure 1**).

Figure 1. Proportion of Patients Currently Not Experiencing a Flare

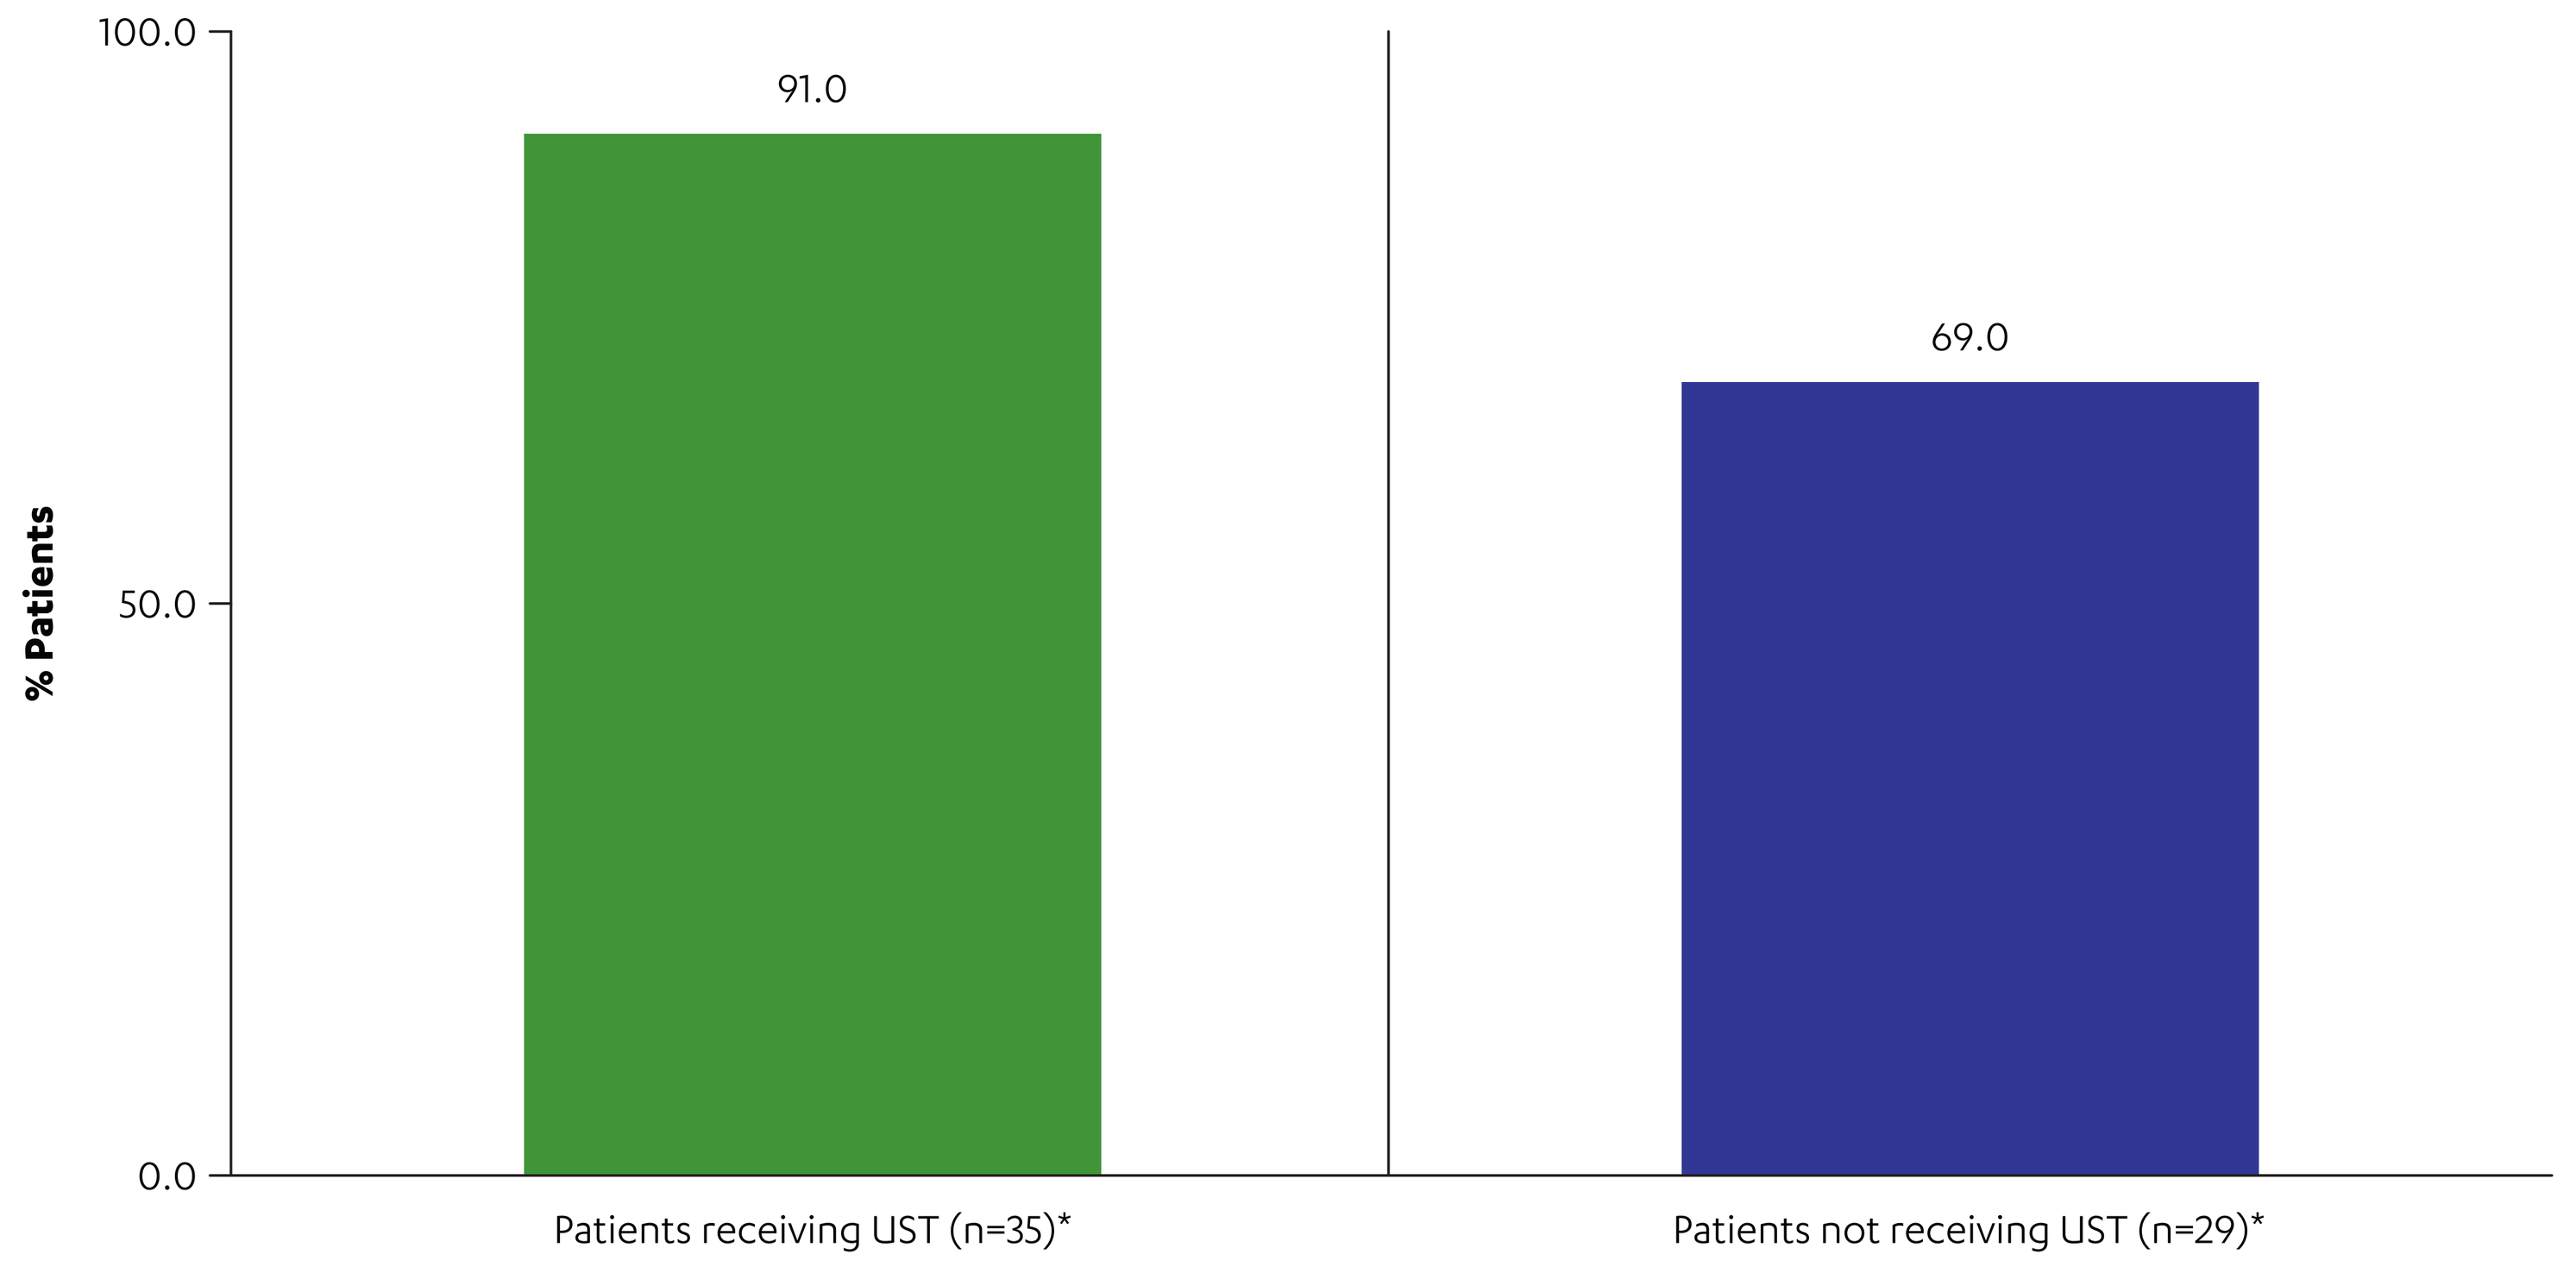
100.0
50.0
0.0
% Patients
91.0
69.0
Patients receiving UST (n=35)*
Patients not receiving UST (n=29)*

*Patients with known flaring status

- Abdominal pain/cramps and non-bloody diarrhea were the most prevalent current symptoms amongst both UST and non-UST groups.
 - In the UST group, 69.2% and 43.6% had these two symptoms, respectively; while among patients not receiving UST, these figures were 87.2% and 71.8%, respectively.
- The mean total SIBDQ score was 52.8 for the UST patient group and 35.5 for the non-UST group (**Figure 2**).
 - A 9-point change is considered a clinically meaningful difference for the SIBDQ[2].
 - The UST patient group had numerically higher domain scores in all four domains vs. the non-UST patient group (**Figure 2**).

Figure 2. Mean Total SIBDQ Scores and Domain Scores (Converted)

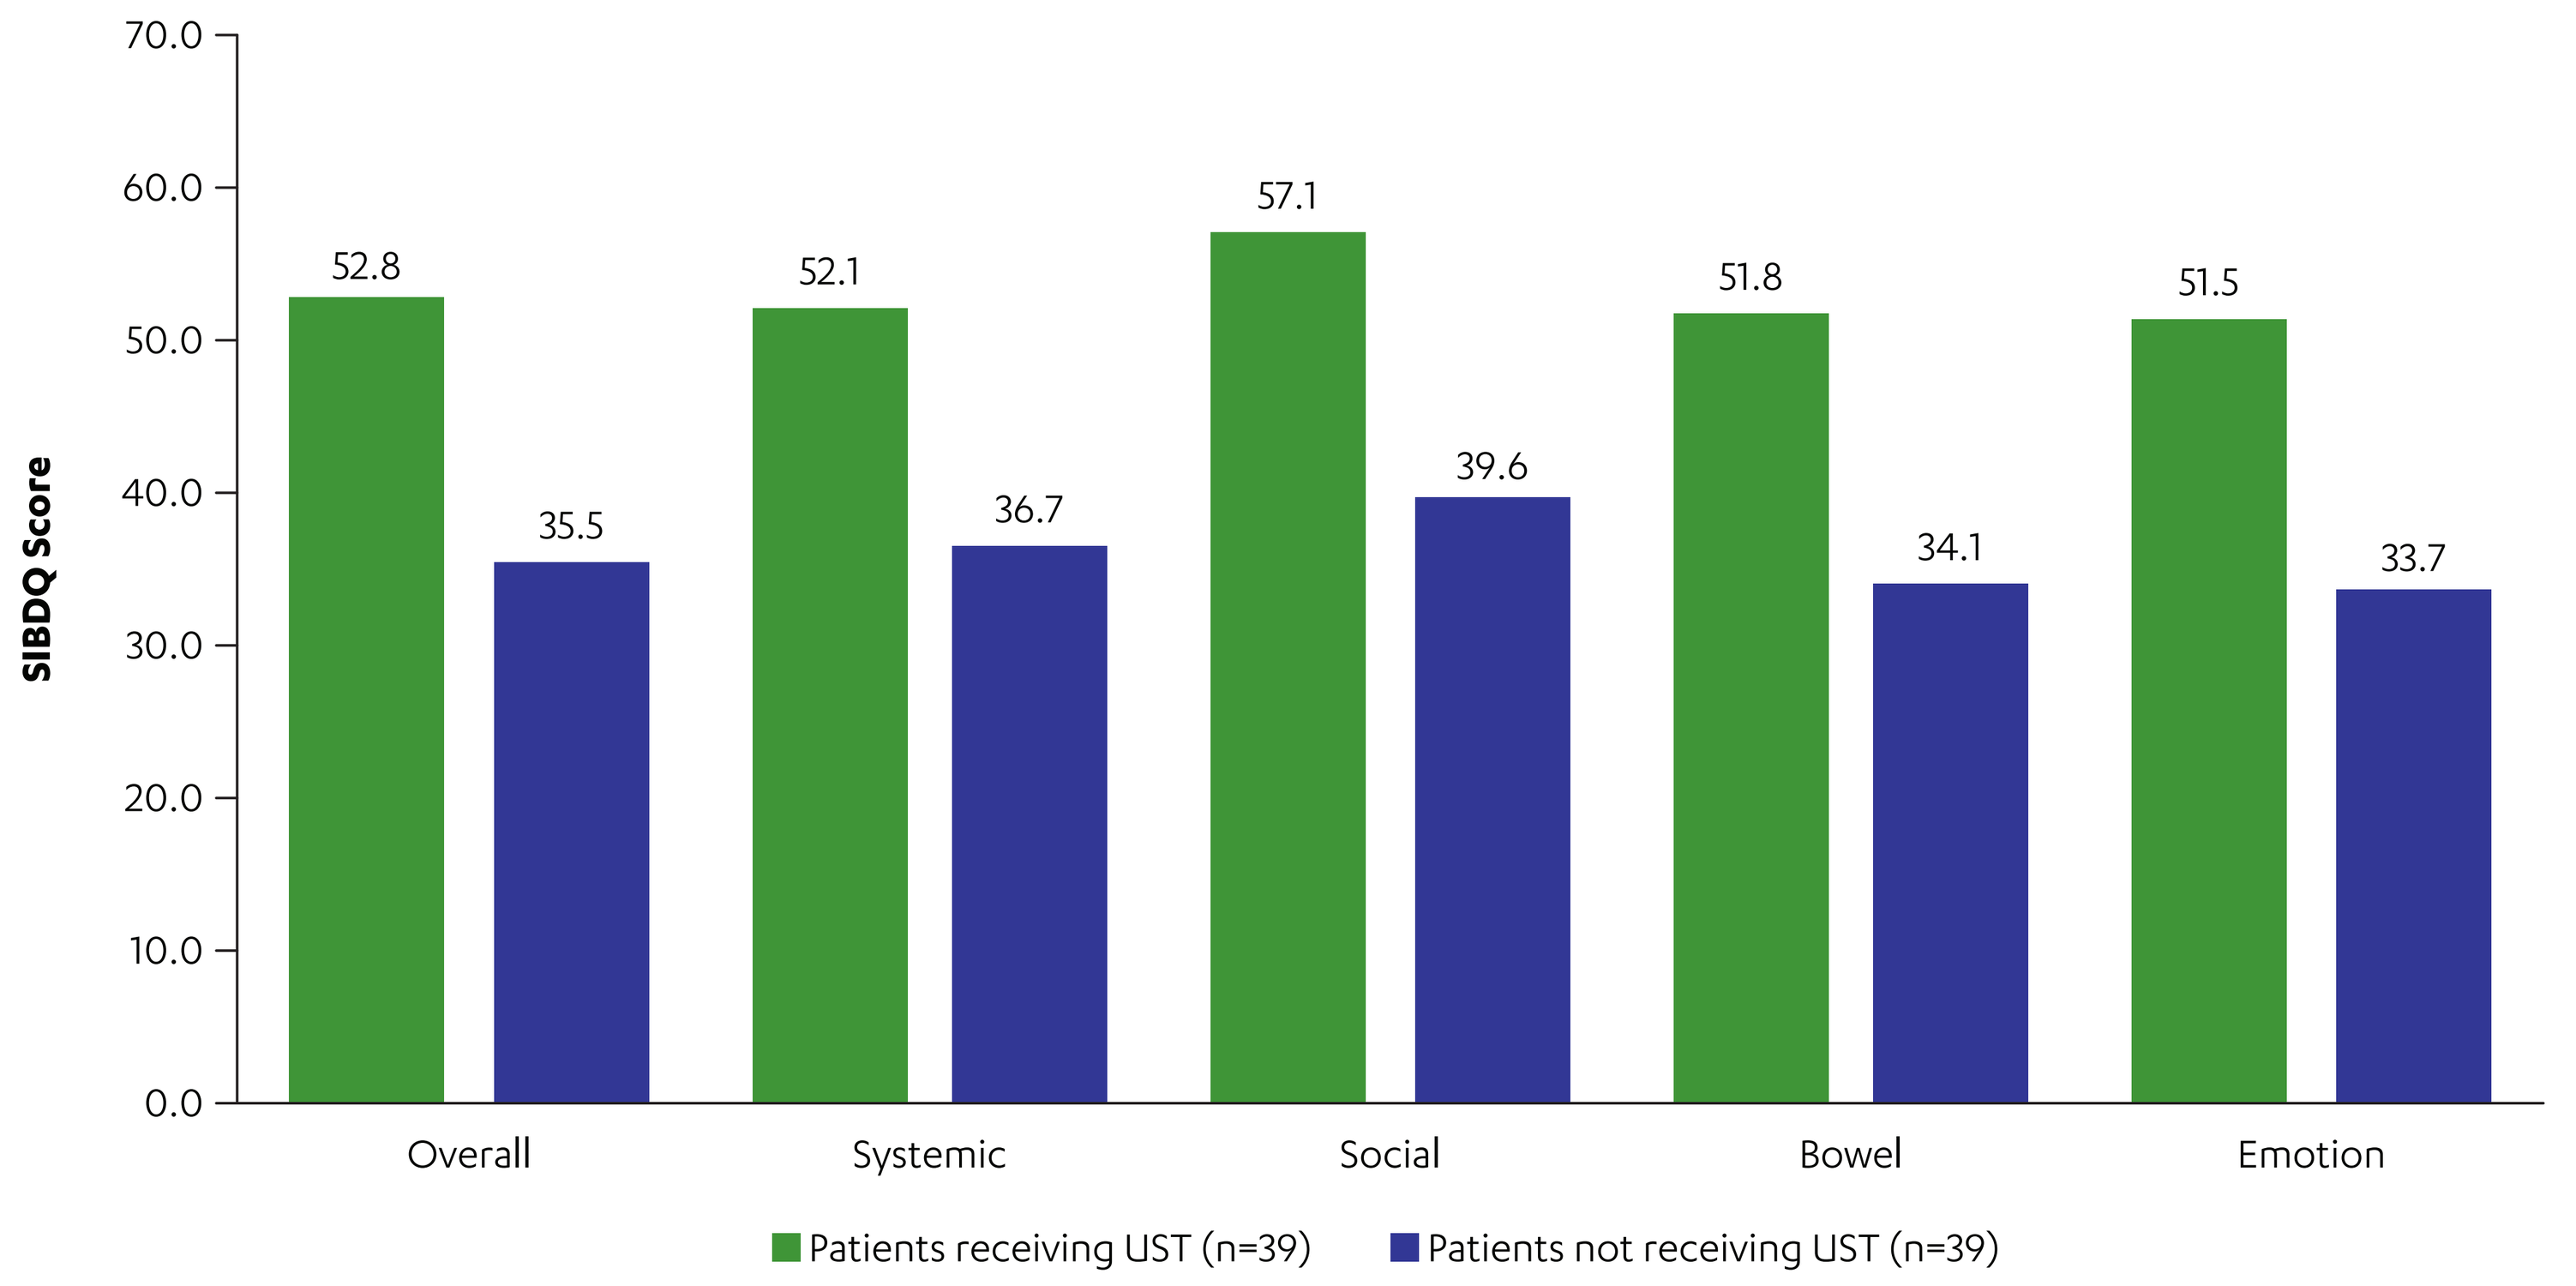
70.0
60.0
50.0
40.0
30.0
20.0
10.0
0.0
SIBDQ Score
52.8
35.5
52.1
36.7
57.1
39.6
51.8
34.1
51.5
33.7
Overall
Systemic
Social
Bowel
Emotion
Patients receiving UST (n=39)
Patients not receiving UST (n=39)

All domain scales have been converted to a 10-70 scale

- An EQ-5D score of 0.91 was reported for the UST patient group, and 0.75 for the non-UST group. The clinically meaningful difference between EQ-5D scores is 0.074[6] (**Figure 3**).

Figure 3. Mean EQ-5D Scores

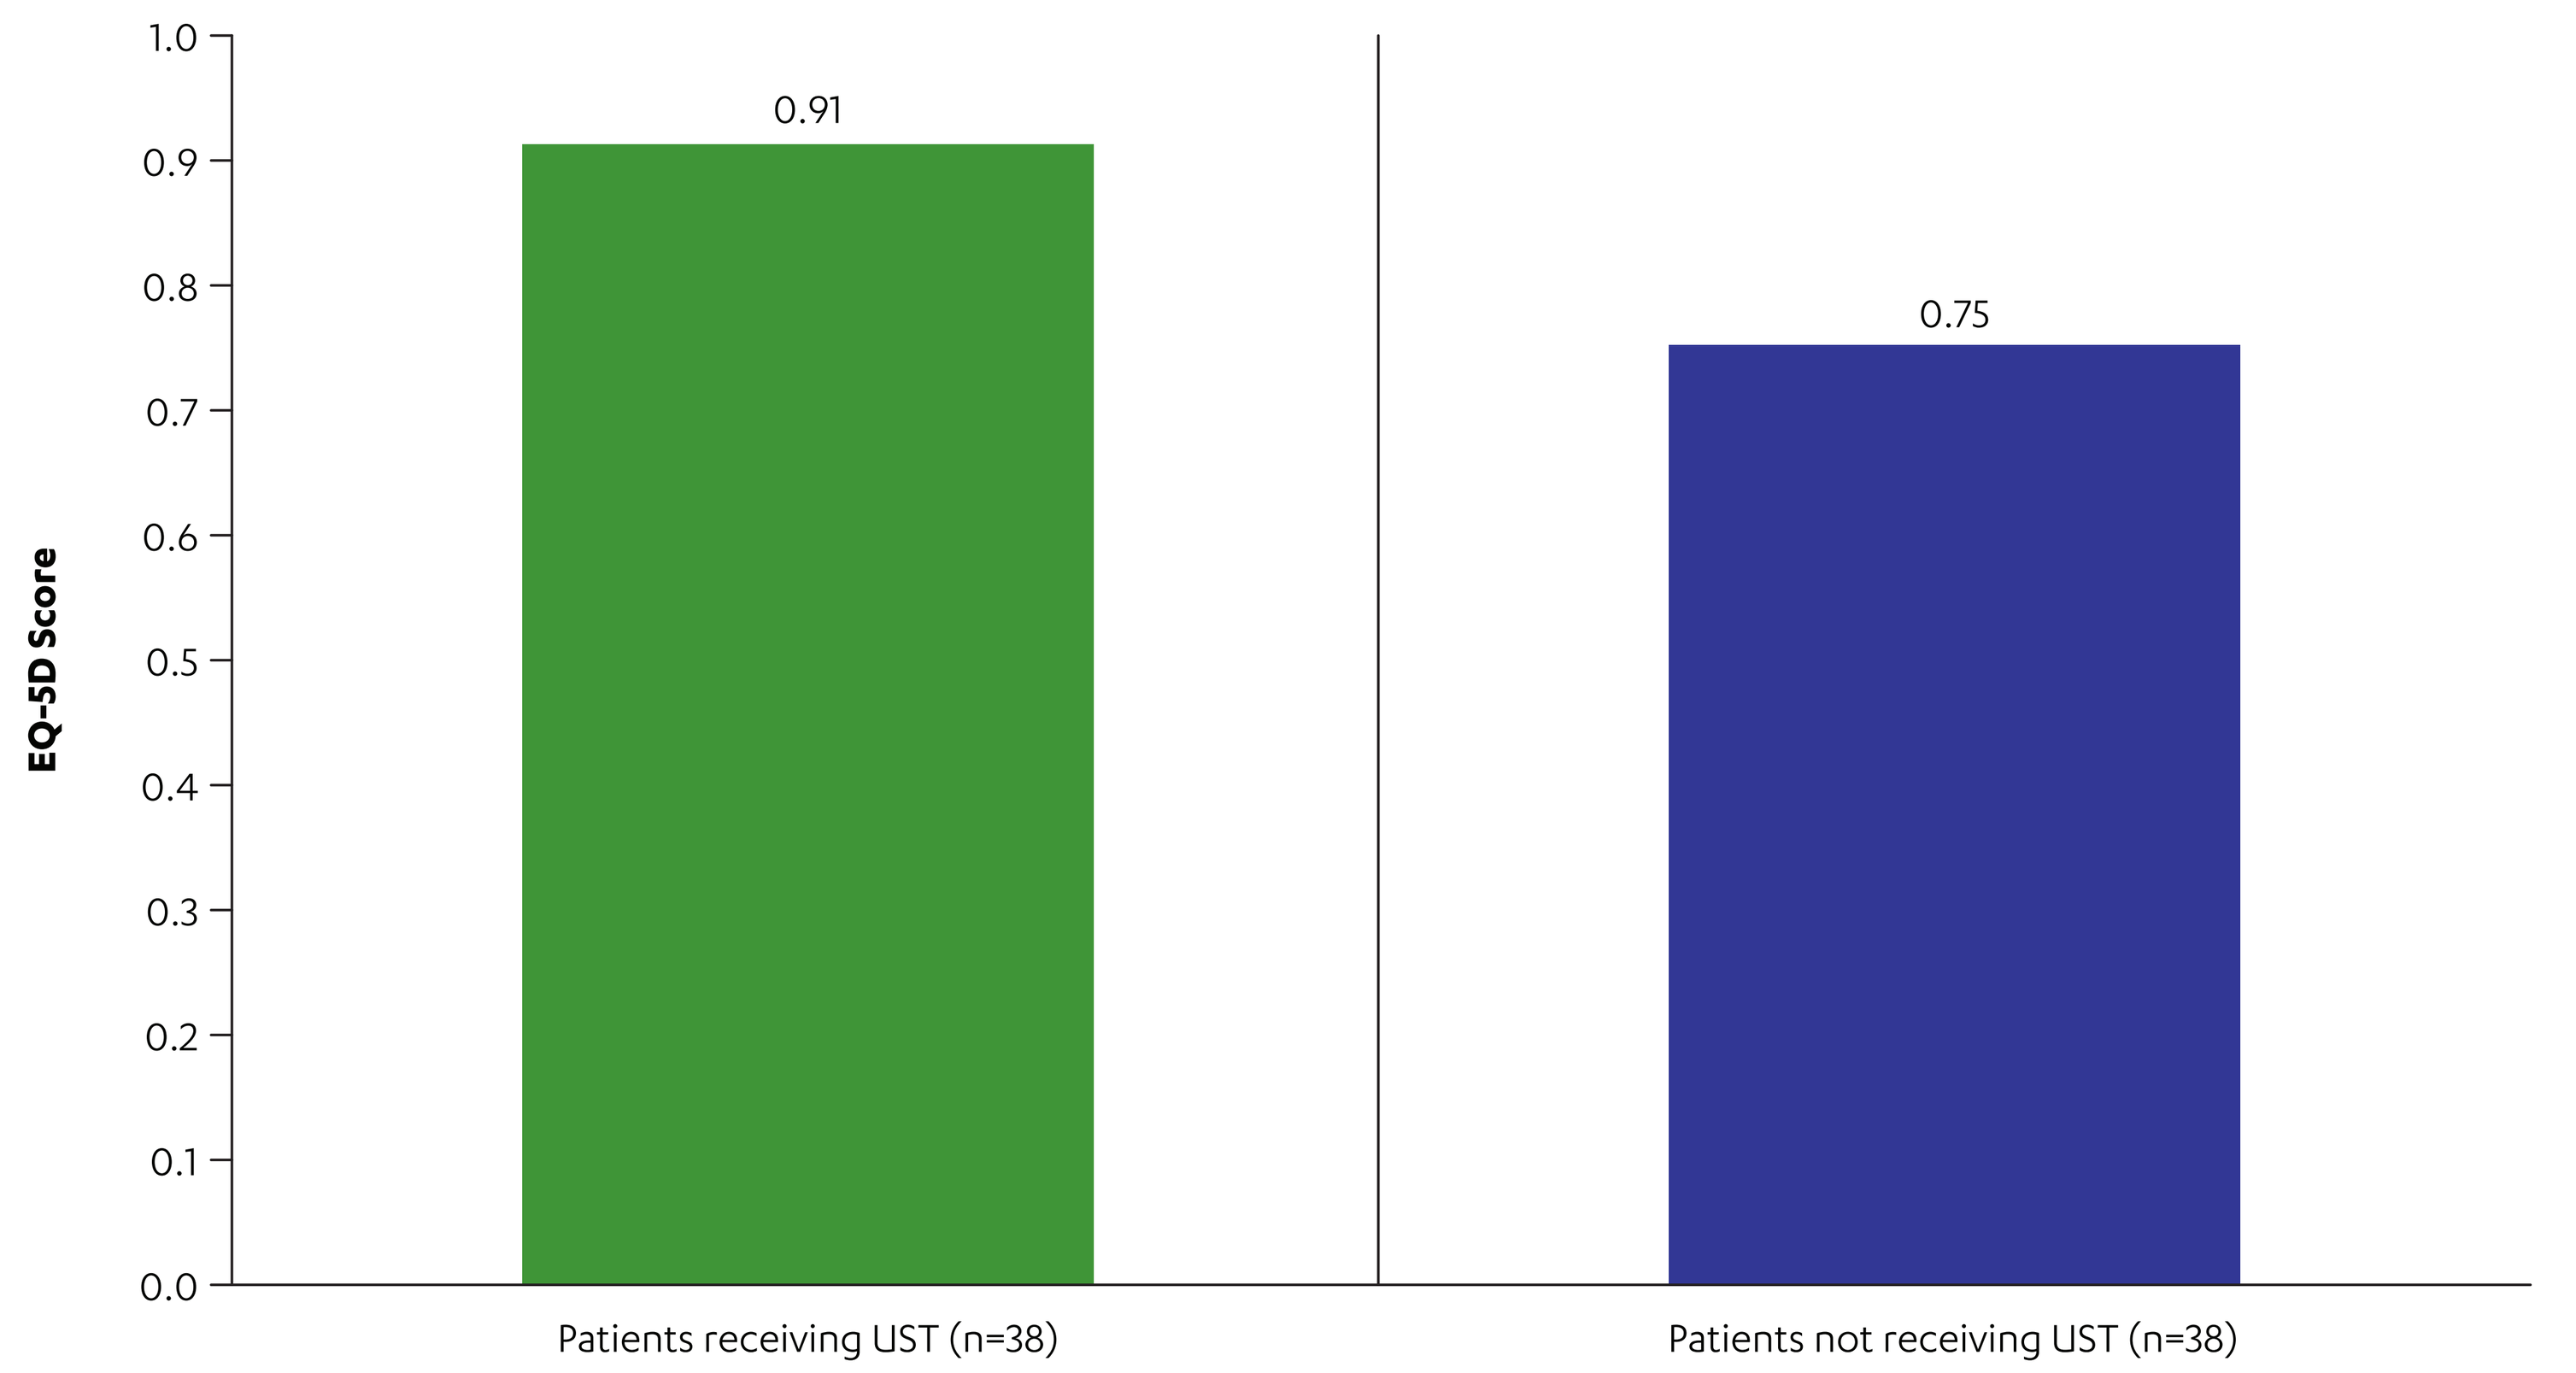
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0.0
EQ-5D Score
0.91
0.75
Patients receiving UST (n=38)
Patients not receiving UST (n=38)

Only includes patients who have completed the EQ-5D component of the PSC

- Mean overall work impairment at last consultation was 21.1% in the UST patient group and 51.0% for the non-UST group. At last consultation, UST and non-UST patients had mean presenteeism of 19.8% and 44.9%, and mean absenteeism of 3.2% and 16.7%, respectively (**Figure 4**).

Figure 4. Absenteeism, Presenteeism, and Total Work Productivity Loss Percentage Impairment, Mean (SD)

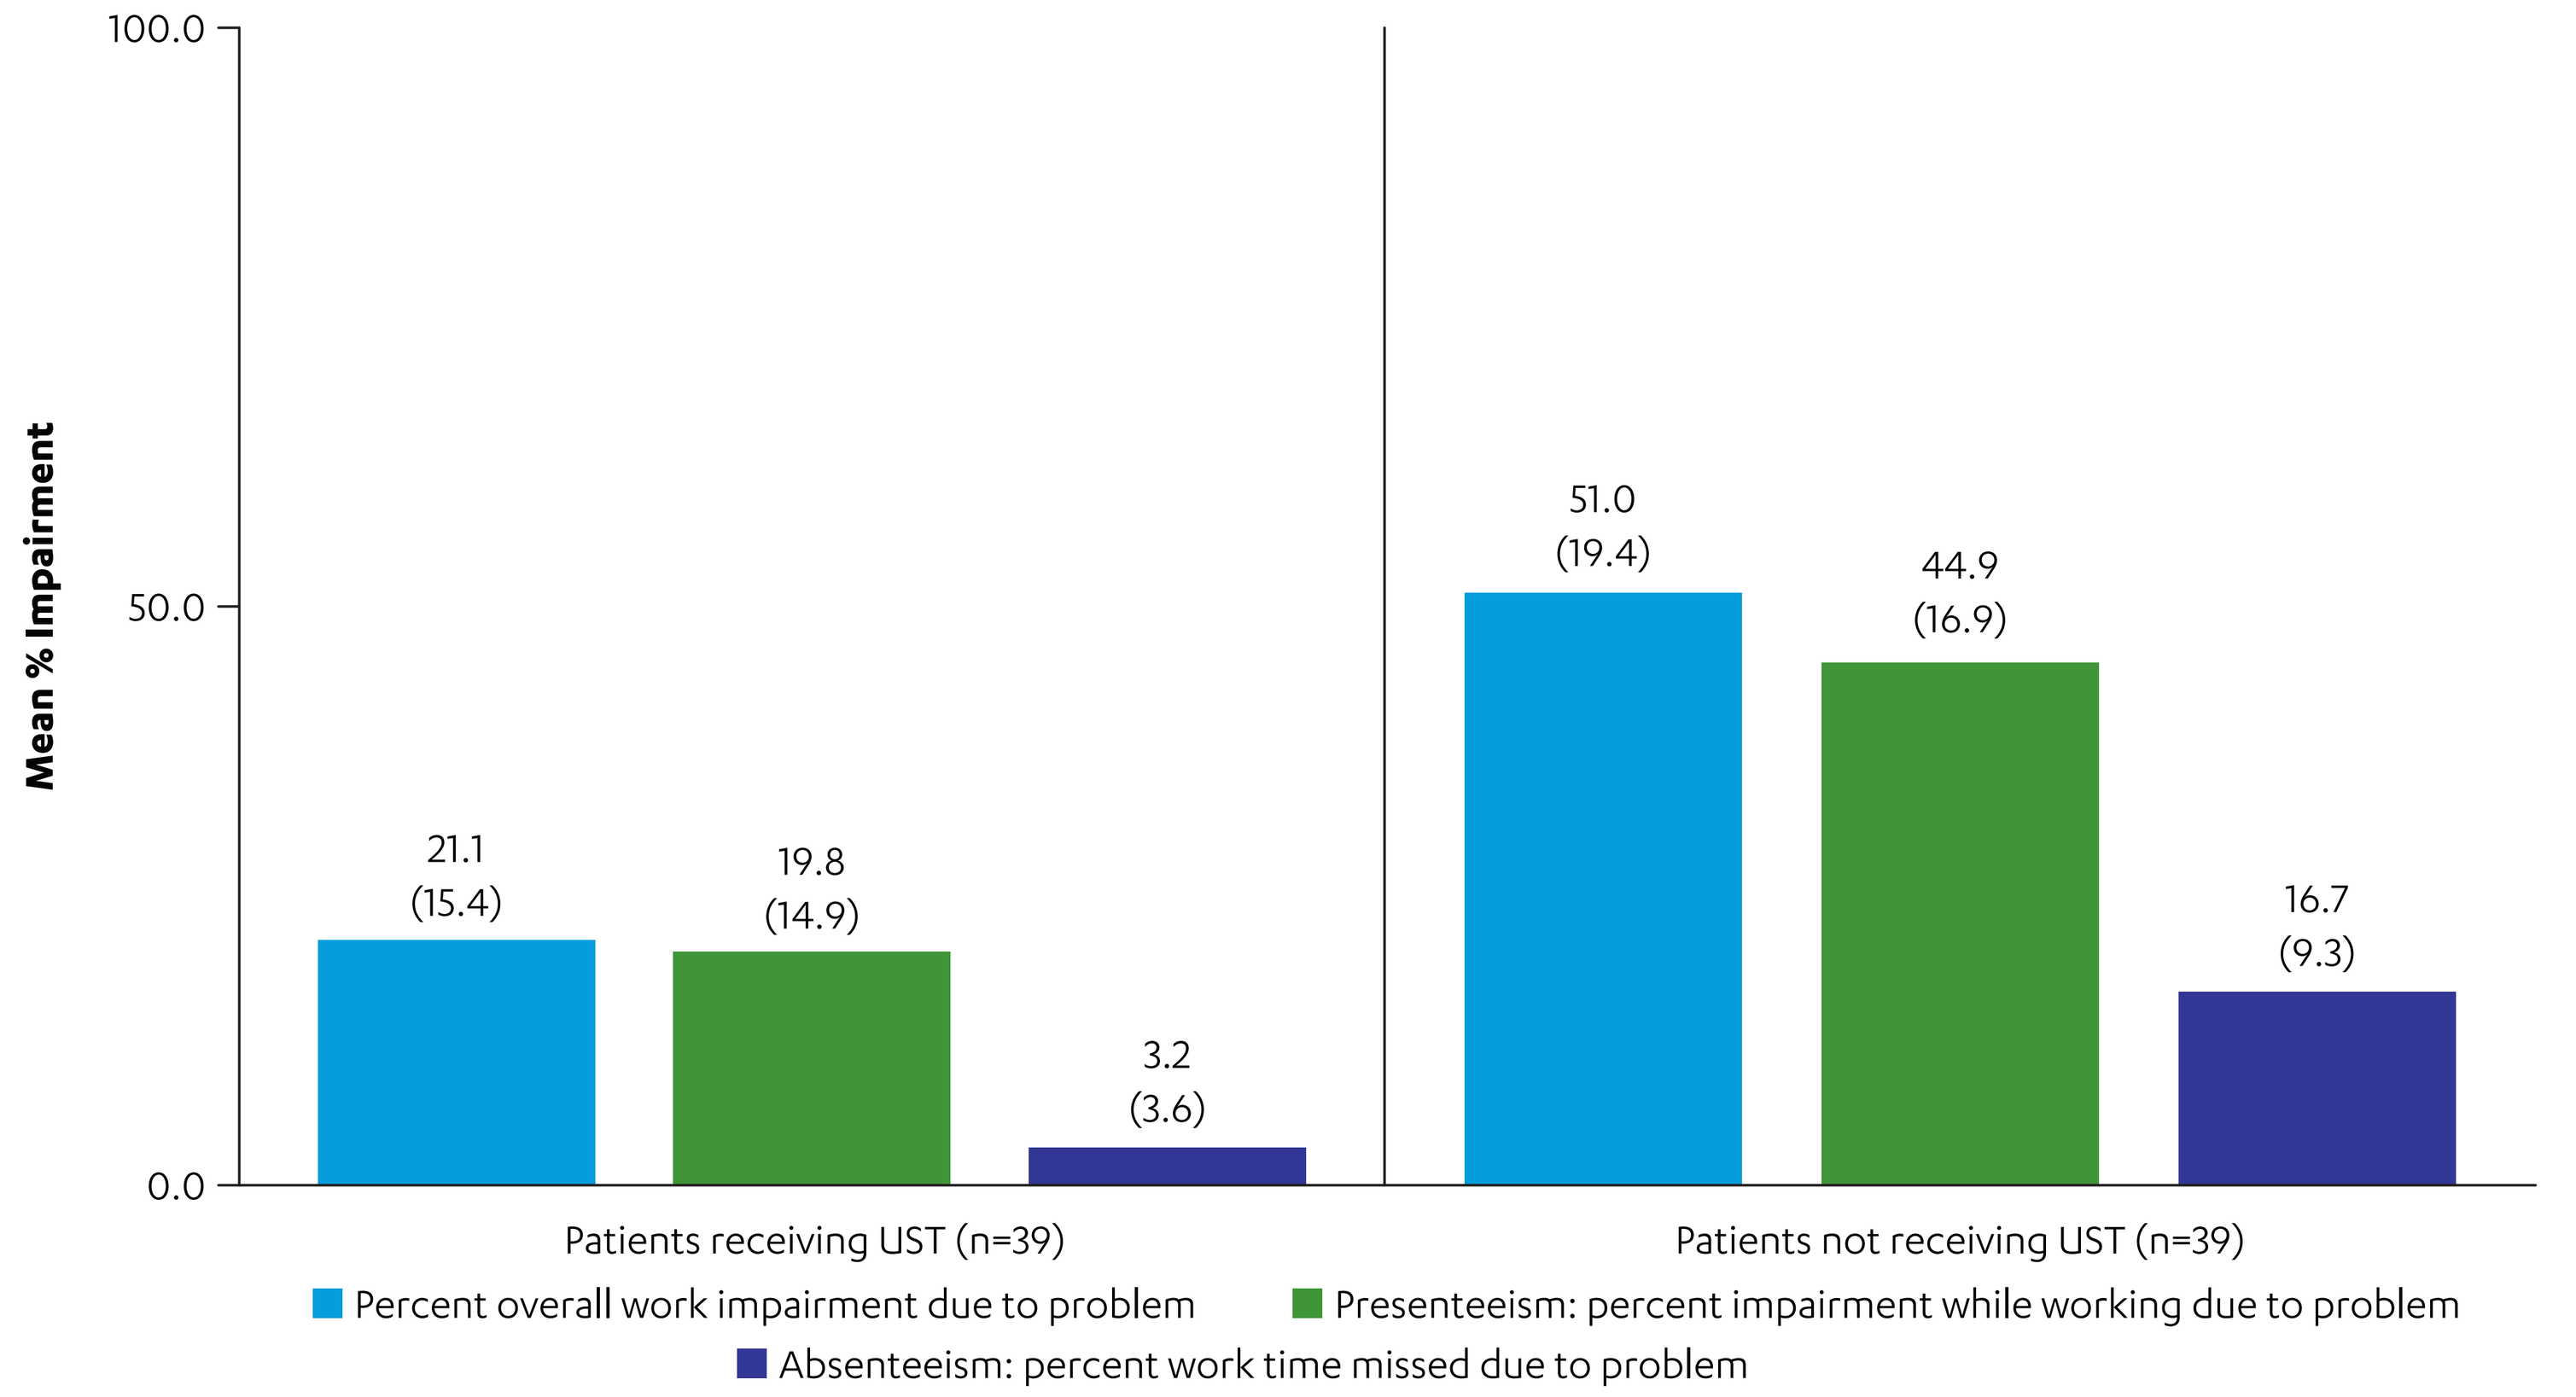
100.0
50.0
0.0
Mean % Impairment
21.1
(15.4)
19.8
(14.9)
3.2
(3.6)
51.0
(19.4)
44.9
(16.9)
16.7
(9.3)
Patients receiving UST (n=39)
Patients not receiving UST (n=39)
Percent overall work impairment due to problem
Presenteeism: percent impairment while working due to problem
Absenteeism: percent work time missed due to problem

- The associated mean annual total WPL costs per patient were $12,211 for the UST patient group and $30,096 for the non-UST group (**Figure 5**).

Figure 5. Mean Costs Associated With Absenteeism, Presenteeism, and Total Work Productivity Loss

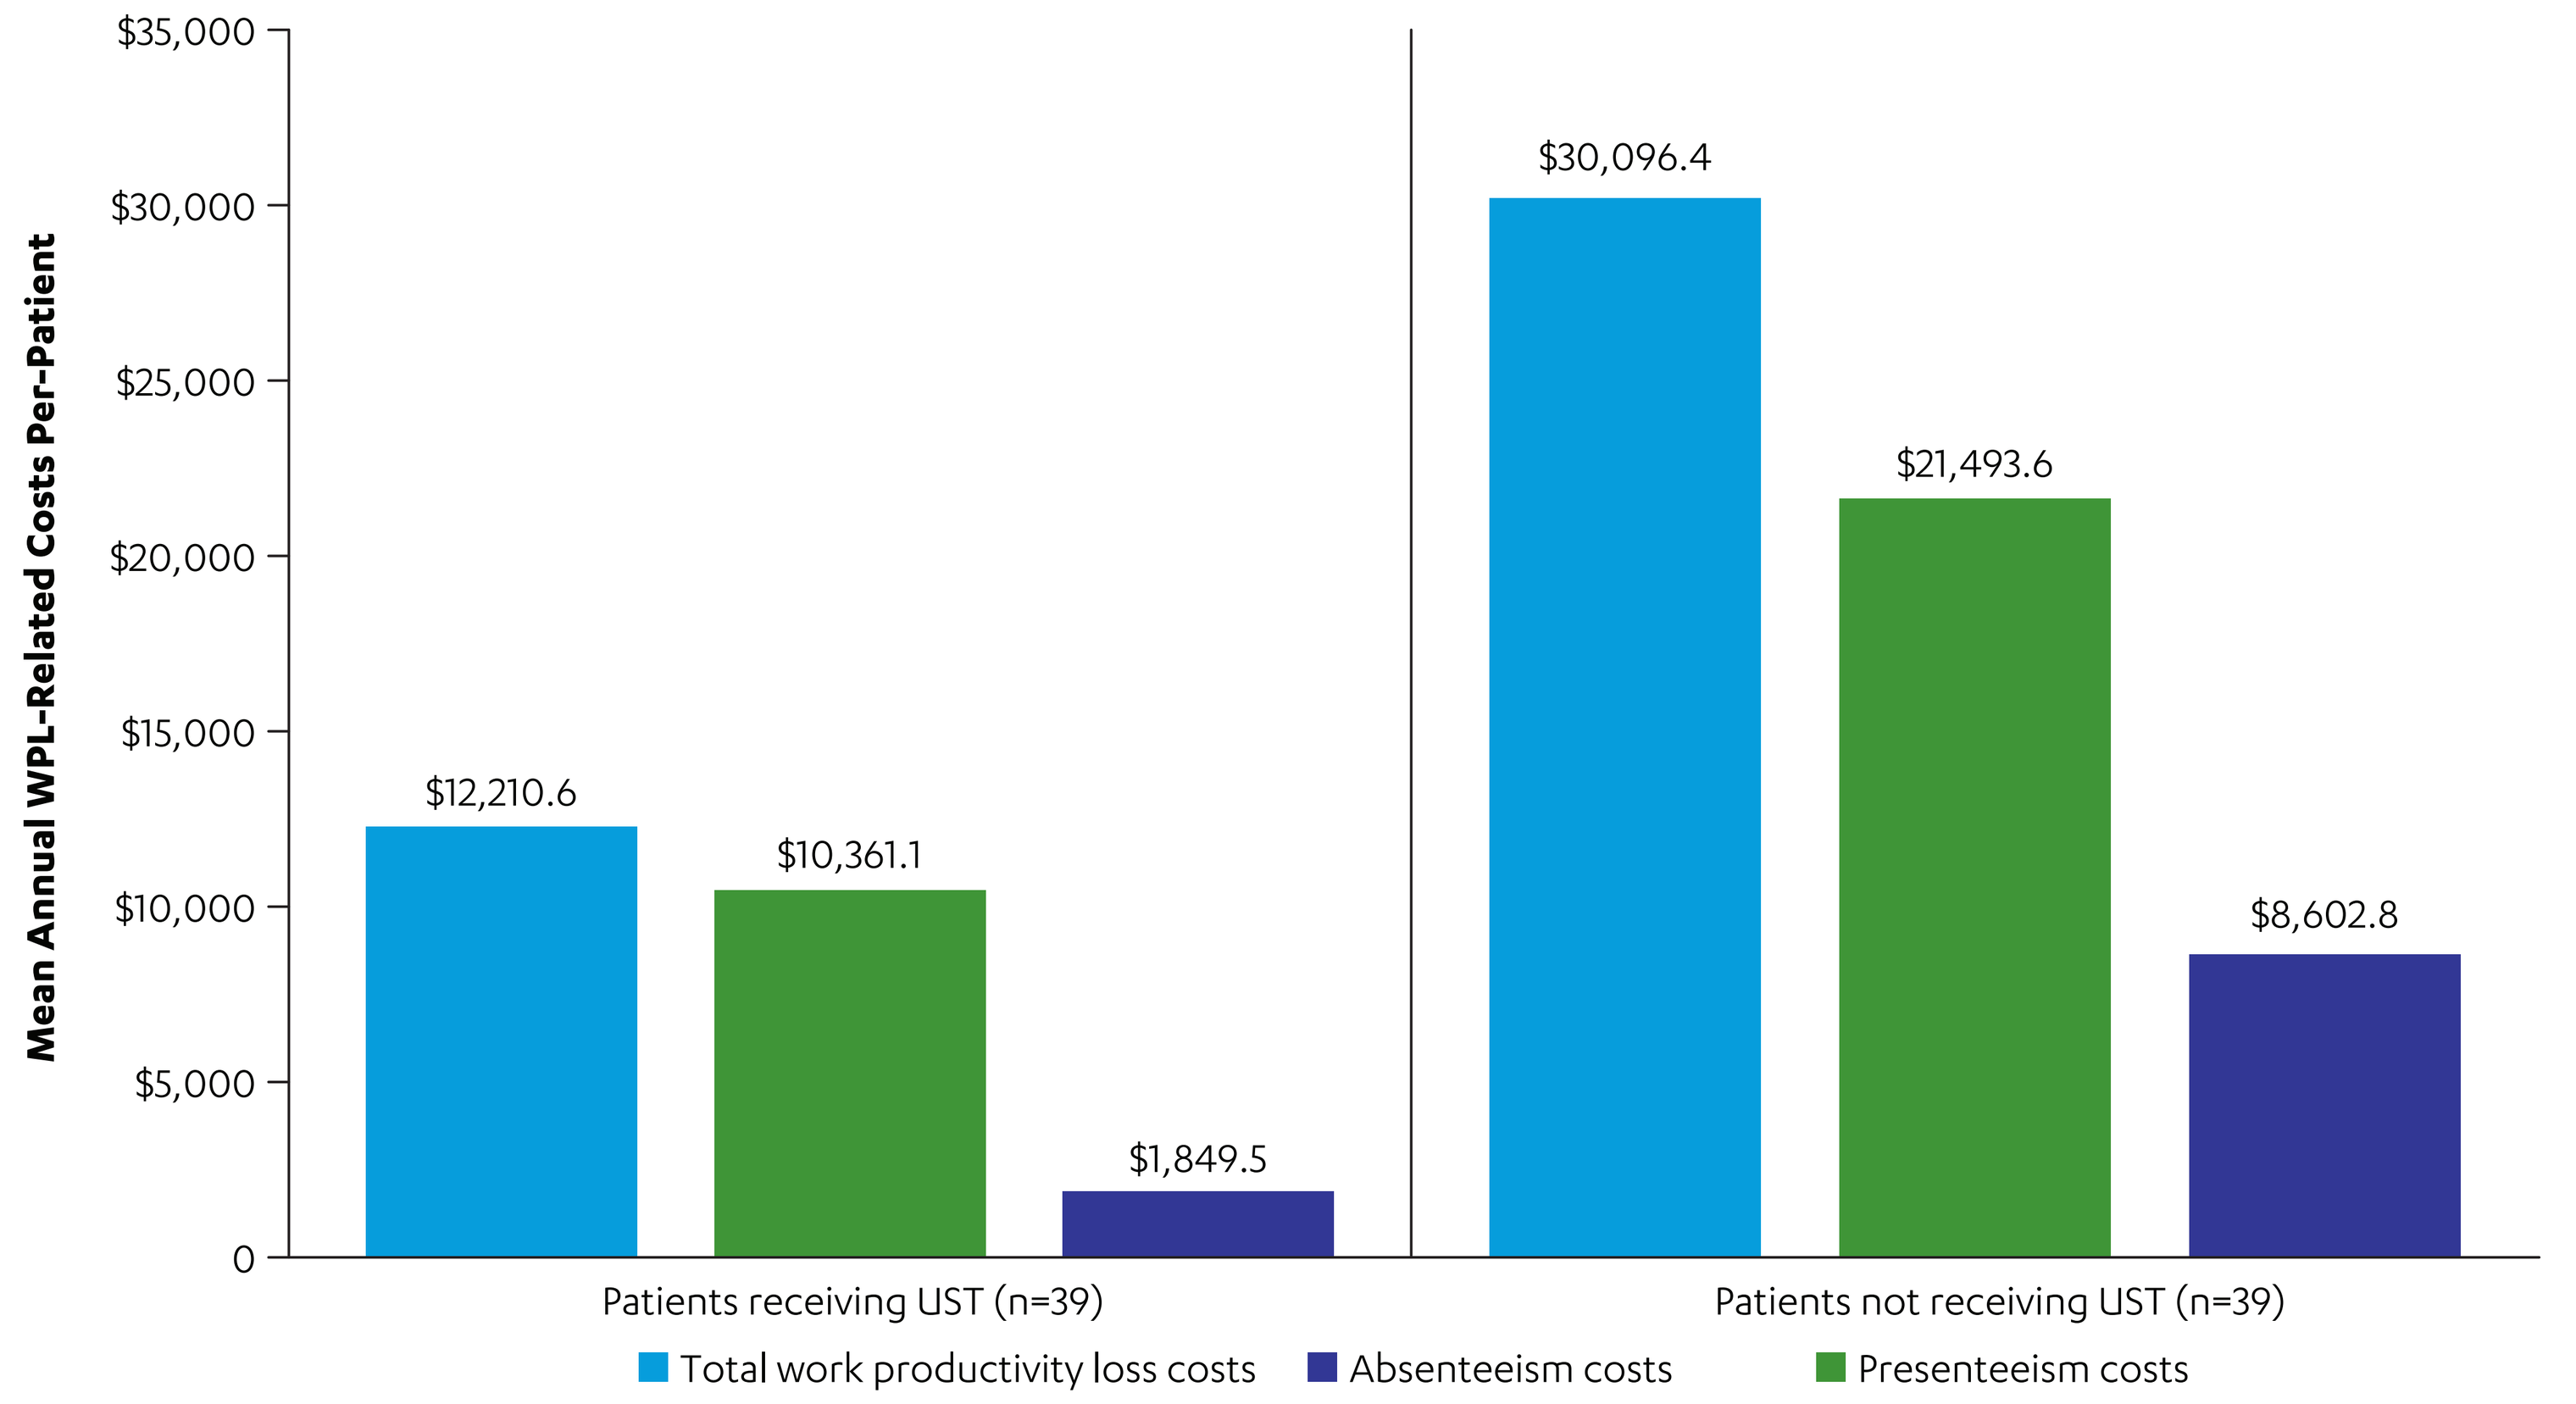
$35,000
$30,000
$25,000
$20,000
$15,000
$10,000
$5,000
0
Mean Annual WPL-Related Costs Per-Patient
$12,210.6
$10,361.1
$1,849.5
$30,096.4
$21,493.6
$8,602.8
Patients receiving UST (n=39)
Patients not receiving UST (n=39)
Total work productivity loss costs
Absenteeism costs
Presenteeism costs

STRENGTHS

- The DSP™ methodology collects two independent large real-world samples of gastroenterologist consulting CD patients who are either currently receiving UST or currently not receiving UST (in moderate-severe disease state according to CDAI) in the US.

LIMITATIONS

- Patients participating in the surveys may not reflect the general CD UST population as patients are visiting their physician and are those who visit more frequently and may be more severely affected than those who do not consult their physician as frequently.
- The quality of data may be subject to recall bias, as this analysis was based on patient and physician survey data.

CONCLUSIONS

- Real-world effectiveness of UST in CD patients was evident from reduced absenteeism, presenteeism and costs associated with WPL following UST initiation, thereby reducing the overall economic burden associated with CD.
- CD Patients treated with UST had numerically higher quality of life vs. CD patients who were not receiving UST.
- Findings from this analysis in the real world are consistent with results from clinical trials of ustekinumab.[7,8]

REFERENCES

1. Anderson P, et al. *Curr Med Res Opin*. 2008;24(11):3063-72
2. Irvine EJ, et al., *Am J Gastroenterol*. 1996;91(8):1571-8.
3. EuroQol Group., *Health Policy*. 1990;16(3):199-208.
4. Reilly MC, Zbrozek AS, Dukes EM. The validity and reproducibility of a work productivity and activity impairment instrument. *Pharmacoeconomics*. 1993; 4: 353–65.
5. U.S. Census Bureau 2019
6. Walters, S.J., Brazier, J.E. Comparison of the minimally important difference for two health state utility measures: EQ-5D and SF-6D. *Qual Life Res* 14, 1523–1532 (2005).
7. Ding Z, et al. Improvements In Work Productivity Loss And Associated Cost Reductions With Ustekinumab And Adalimumab In Biologic-Naïve Patients With Moderately-To-Severely Active Crohn's Disease: Results From The SEAVUE Study. 2021. *Am. J. Gastroenterol. Suppl.*
8. Danese S, et al. Health-related quality of life with ustekinumab versus adalimumab for induction and maintenance therapy in biologic-naive patients with moderate-to-severe Crohn's disease: IBDQ in the SEAVUE study. 2021. *J. United Eur Gastroent. Suppl.*

Presented at the 27th Annual Meeting of the International Society for Pharmacoeconomics and Outcomes Research May 15–18, 2022; National Harbor, Washington, DC

This study was supported by Janssen Scientific Affairs, LLC