EE281

Evaluation of Health Care Resource Utilization and Costs Among Bio-Naive Crohn's Disease Patients Initiating Biologics With Two Years of Follow-Up

A. Cyhaniuk[1], R. Zhao[2], Z. Ding[2], P. Gupta[1], S. Kachroo[2]

[1]STATinMED Research, Ann Arbor, MI; [2]Janssen Scientific Affairs, LLC, Horsham, PA

Scan the QR code

https://www.congresshub.com/Immunology/ISPOR2022/Ustekinumab/Zhao-Bio-Naive

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

BACKGROUND

- Crohn's disease (CD) is a relapsing chronic inflammatory disorder of the gastrointestinal tract which may involve any part of the gastrointestinal tract from the mouth to the anus.[1,2] Its treatment incurs high health care resource utilization and costs in CD patients.[3,4]
- There is a need for real-world evidence on health care resource utilization (HCRU) and costs among bio-naive CD patients over a 2-year follow-up period.

OBJECTIVE

This study examined Crohn's Disease (CD)-related health care resource utilization (HCRU) and costs among bio-naïve CD patients initiating biologics over a 2-year follow-up period. We examined CD patients who initiated adalimumab (ADA), certolizumab pegol (CZP), infliximab (IFX), ustekinumab (UST), and vedolizumab (VDZ).

METHODS

- Databases: The study used claims data pooled from three US commercial databases: IQVIA PharMetrics™, IBM® MarketScan®, and Optum Clinformatics.
- Study Period:
 September 26, 2015 – February 29, 2020 (For Optum Clinformatics and for IQVIA PharMetrics)
 September 26, 2015 – September 30, 2019 (For IBM MarketScan)
- Identification Period:
 September 26, 2016 – February 29, 2018 (For Optum Clinformatics and for IQVIA PharMetrics)
 September 26, 2016 – September 30, 2017 (For IBM MarketScan)
- Index Date: Index date was defined as the first claim date for ADA, CZP, IFX, UST, or VDZ among adult patients with ≥1 CD diagnosis during the identification period and without any prior biologic usage in the baseline period. Patients could be assigned to multiple cohorts if they used more than one biologic during the identification period, i.e., study cohorts were not mutually exclusive. CZP was excluded from further analysis due to small sample size.
- Baseline Period: 1-year prior to the index date
- Follow-up Period: 2-year post the index date

Figure 1. Patient Selection Criteria

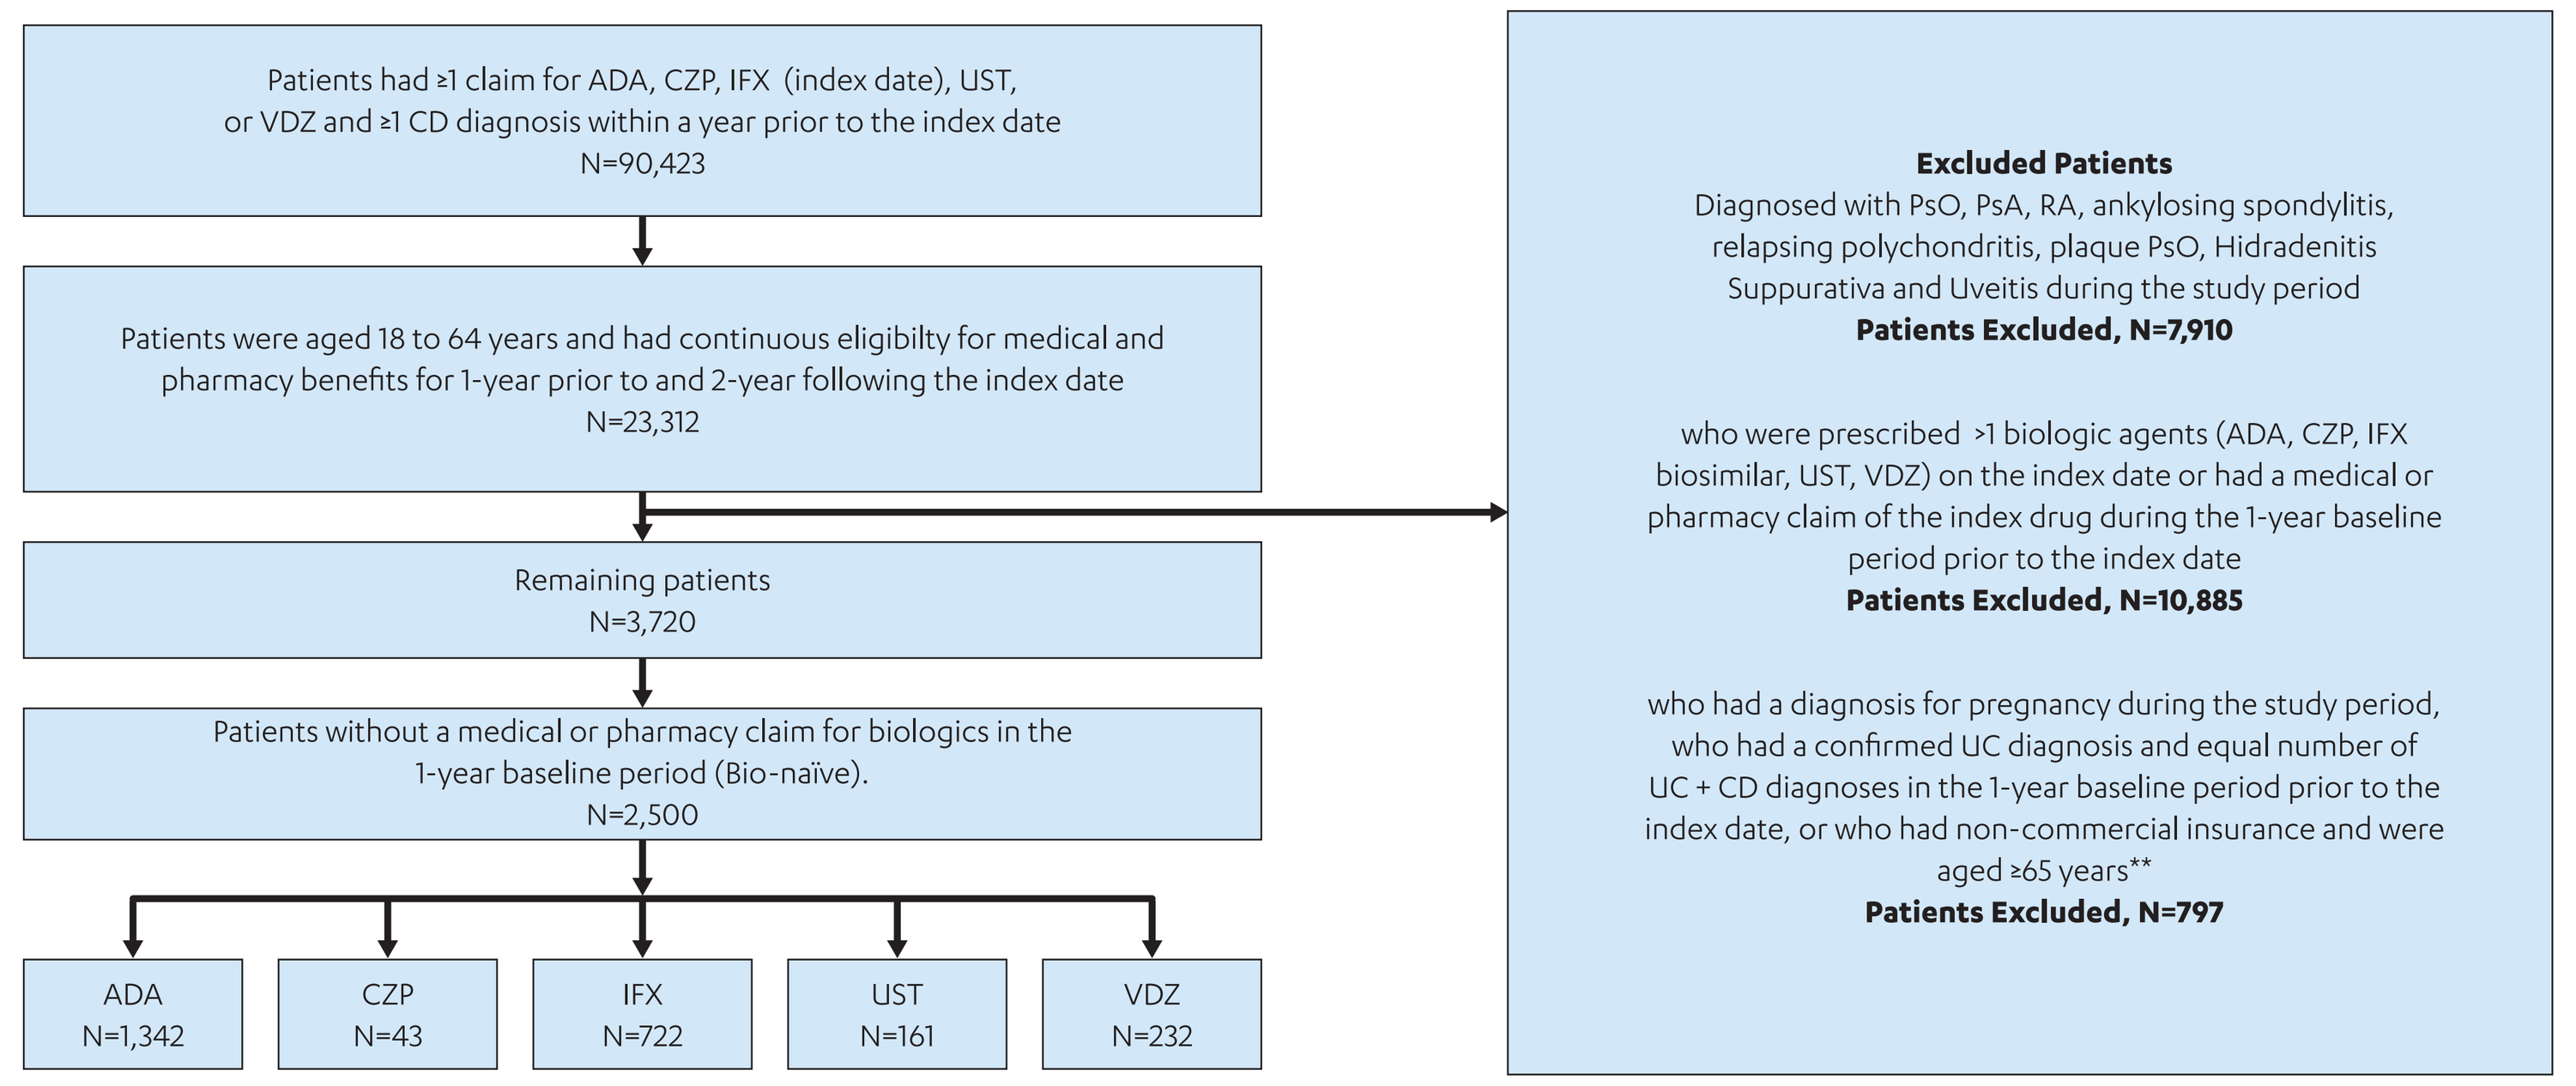
Patients had ≥1 claim for ADA, CZP, IFX (index date), UST, or VDZ and ≥1 CD diagnosis within a year prior to the index date
N=90,423
Patients were aged 18 to 64 years and had continuous eligibilty for medical and pharmacy benefits for 1-year prior to and 2-year following the index date
N=23,312
Remaining patients
N=3,720
Patients without a medical or pharmacy claim for biologics in the 1-year baseline period (Bio-naïve).
N=2,500
ADA N=1,342
CZP N=43
IFX N=722
UST N=161
VDZ N=232
Excluded Patients
Diagnosed with PsO, PsA, RA, ankylosing spondylitis, relapsing polychondritis, plaque PsO, Hidradenitis Suppurativa and Uveitis during the study period
Patients Excluded, N=7,910
who were prescribed >1 biologic agents (ADA, CZP, IFX biosimilar, UST, VDZ) on the index date or had a medical or pharmacy claim of the index drug during the 1-year baseline period prior to the index date
Patients Excluded, N=10,885
who had a diagnosis for pregnancy during the study period, who had a confirmed UC diagnosis and equal number of UC + CD diagnoses in the 1-year baseline period prior to the index date, or who had non-commercial insurance and were aged ≥65 years**
Patients Excluded, N=797

ADA: adalimumab; CD: Crohn's disease; CZP: certolizumab pegol; IFX: infliximab; PsA: psoriatic arthritis; RA: rheumatoid arthritis; UC; ulcerative colitis; UST: ustekinumab; VDZ: vedolizumab
*Patients could be assigned to multiple cohorts based on the biologics used during the identification period.
**A modified algorithm based on the Manitoba study was used to exclude patients with an ulcerative colitis (UC) diagnosis or those with equal number of UC and CD diagnosis.[5,6]

Baseline and Outcome Variables

- Baseline variables: age, sex, US geographic region, Charlson comorbidity index (CCI) score, baseline comorbidities.
- Outcome variables: health care resource utilization (HCRU) and associated costs in the second year of follow-up period were evaluated, including emergency room (ER) visits, surgery claims, and inpatient stays. CD-related (defined as claims associated with a CD diagnosis, treatment for CD and its comorbid conditions) and all-cause HCRU and associated costs were reported.

Statistical Analysis

- Descriptive analyses were conducted in the pooled population. Means and standard deviations were computed for continuous variables, and frequency and percentages were calculated for categorical variables.
- The percentage change in CD-related hospitalization, ER visits, surgeries and associated costs were calculated.

Relative change from baseline=(Utilization or cost during follow up - Utilization or cost during baseline)/Utilization or cost during baseline period

- ***Pre- vs Post-Index Analysis:*** In each study biologic cohort, CD-related and all-cause HCRU and costs were compared between the baseline and the second year of follow-up period using a paired two-sample t-test (for continuous variables) or McNemar's test (for categorical variables). A p-value of <0.05 was considered statistically significant.
- Costs were adjusted to 2019 US dollars.

RESULTS

After applying all selection criteria (**Figure 1**), bio-naive patients were selected as follows: 1,342 ADA, 43 CZP, 722 IFX, 161 UST, 232 VDZ patients. CZP patients were not included in the analyses because of small sample size.

- Patients on UST and VDZ were numerically older and had numerically higher CCI score (**Table 1**).
- Selected comorbidities among bio-naïve CD patients included anemia, anxiety, and depression (**Table 1**).

Table 1. Descriptive Baseline Characteristics

Characteristics	Adalimumab N=1,342	Infliximab N=722	Ustekinumab N=161	Velodizumab N=232
Age, mean	39.1	36.8	40.7	41.9
Age group, in years (%)				
18-34	38.7%	45.6%	32.3%	28.0%
35-54	46.1%	42.5%	52.2%	53.4%
55-64	15.2%	11.9%	15.5%	18.5%
Sex, %				
Male	53.7%	57.3%	51.6%	53.0%
Female	46.3%	42.7%	48.4%	47.0%
US geographic regions, %				
Northeast	19.4%	16.9%	19.3%	16.8%
North Central	30.2%	32.3%	32.3%	33.2%
South	40.2%	39.1%	39.1%	40.9%
West	10.1%	11.4%	9.3%	9.1%
Unknown	0.1%	0.4%	0.0%	0.0%
Charlson comorbidity index score, mean	1.1	1.1	1.2	1.2
Baseline Comorbidities, %				
Anemia	25.9%	31.2%	35.4%	25.4%
Anxiety	18.7%	18.0%	24.8%	15.1%
Antherosclerosis	1.1%	0.1%	0.6%	0.0%
Celiac disease	2.7%	3.3%	3.7%	5.6%
Cholelithiasis	3.8%	3.6%	3.7%	2.6%
Chronic pain	5.6%	4.7%	8.7%	4.7%
Depression	14.8%	16.6%	22.4%	12.9%
Diabetes	5.1%	4.6%	4.3%	6.0%
Fatigue	13.6%	12.6%	14.9%	11.2%
Fistula	6.0%	10.2%	10.6%	4.3%
Obesity	9.8%	9.3%	10.6%	11.2%
Venous Thromboembolism	1.6%	2.1%	3.1%	0.4%

*Due to rounding of the percentages, some of the categorical variables might not add up to a 100%.

Figure 2. Pre-post CD-related and All-cause Hospitalization and Associated Costs Among Bio-naïve Crohn's Disease Patients Initiated Treatment With Biologics

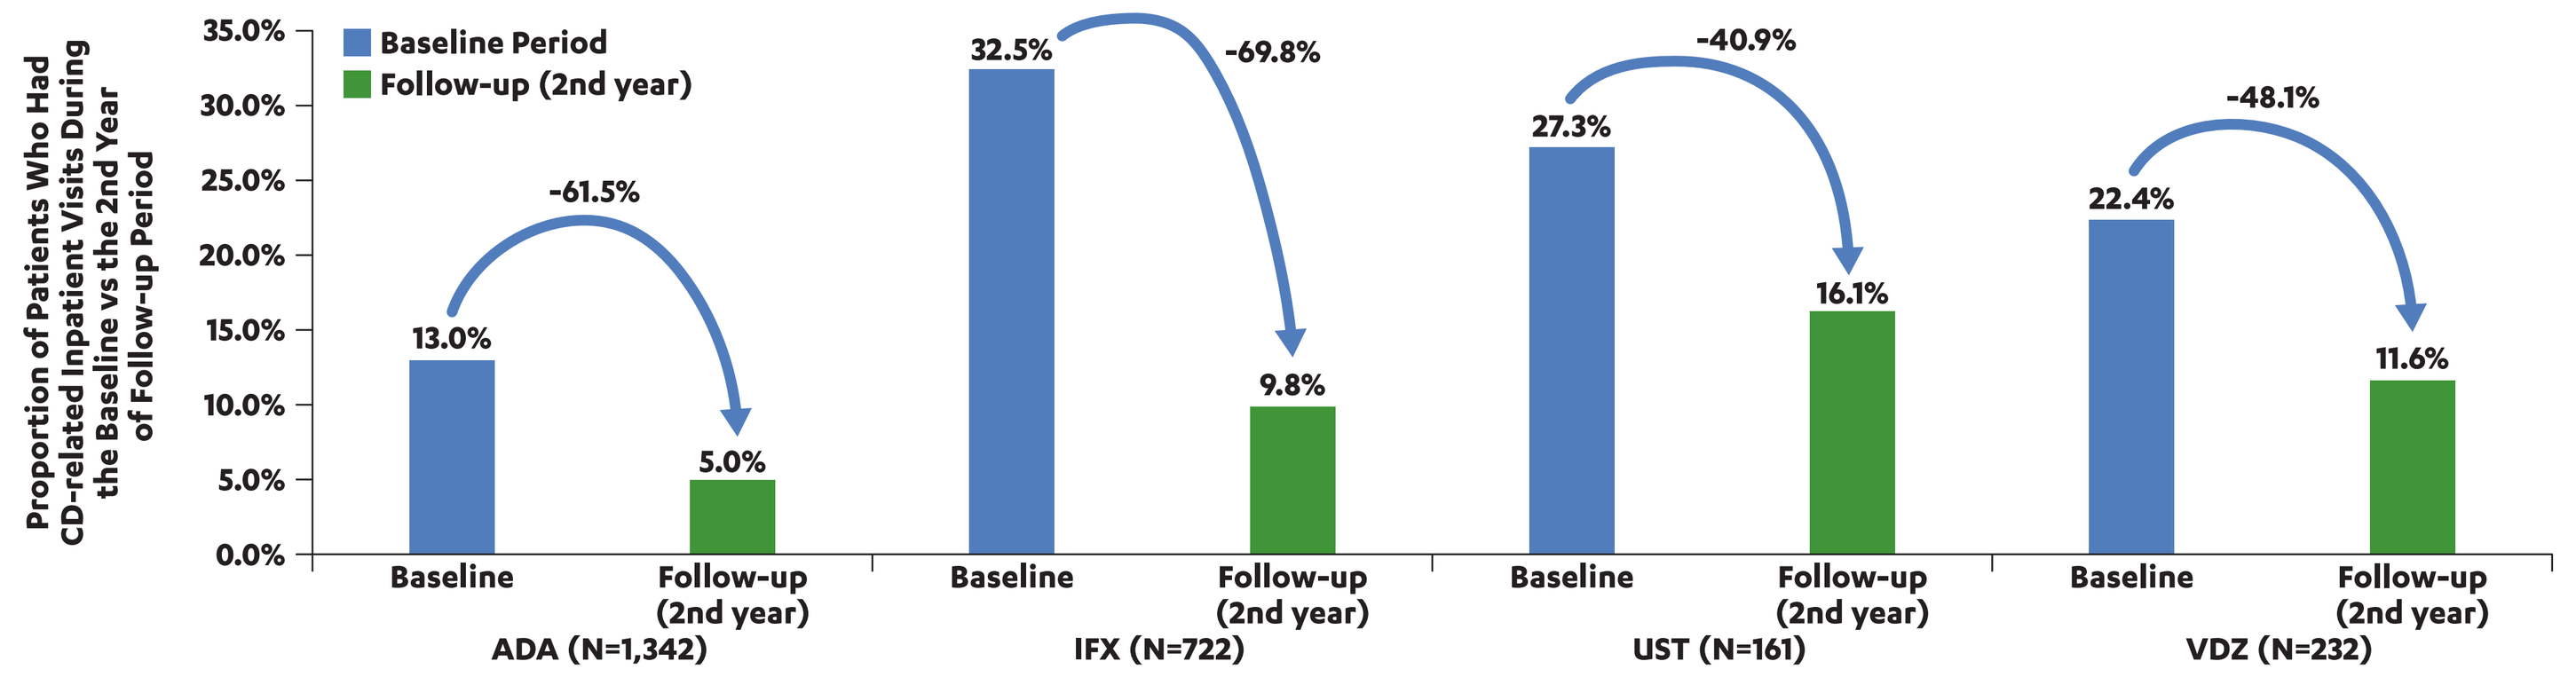
Proportion of Patients Who Had CD-related Inpatient Visits During the Baseline vs the 2nd Year of Follow-up Period
Baseline Period
Follow-up (2nd year)
13.0%
5.0%
-61.5%
32.5%
9.8%
-69.8%
27.3%
16.1%
-40.9%
22.4%
11.6%
-48.1%
Baseline
Follow-up (2nd year)
ADA (N=1,342)
IFX (N=722)
UST (N=161)
VDZ (N=232)

	ADA (N=1,342)		IFX (N=722)		UST (N=161)		VDZ (N=232)	
	Baseline vs Follow-up (2nd year)	Relative change from baseline	Baseline vs Follow-up (2nd year)	Relative change from baseline	Baseline vs Follow-up (2nd year)	Relative change from baseline	Baseline vs Follow-up (2nd year)	Relative change from baseline
CD-related inpatient utilization	13.0% vs 5.0% (p<0.0001)	-61.5%	32.5% vs 9.8% (p<0.0001)	-69.8%	27.3% vs 16.1% (p=0.0422)	-40.9%	22.4% vs 11.6% (p=0.0069)	-48.1%
Mean annual CD-related inpatient costs	$7,460 vs $2,582 (p<0.0001)	-65.4%	$10,402 vs $3,328 (p<0.0001)	-68.0%	$10,271 vs $4,354 (p=0.0163)	-57.6%	$5,565 vs $4,867 (p=0.6837)	-12.5%
All-cause inpatient utilization	14.2% vs 5.4% (p<0.0001)	-61.6%	33.1% vs 10.2% (p<0.0001)	-69.0%	29.2% vs 16.1% (p=0.0192)	-44.7%	23.3% vs 12.9% (p=0.0121)	-44.4%
Mean annual all-cause inpatient costs	$8,589 vs $4,392 (p<0.0001)	-48.9%	$13,177 vs $4,426 (p<0.0001)	-66.4%	$12,218 vs $5,600 (p=0.0216)	-54.2%	$8,062 vs $5,638 (p=0.2757)	-30.1%

ADA: adalimumab; CD: Crohn's disease; IFX: infliximab; UST: Ustekinumab; VDZ: vedolizumab
Relative change from baseline=(Utilization or cost during follow up-Utilization or cost during baseline)/Utilization or cost during baseline period
Note: Statistical significance represented by p-values (p-value <0.05).

- Compared to baseline, proportion of patients with CD-related inpatient stays decreased significantly for ADA (13.0% vs 5.0%; p-value <0.0001), IFX (32.5% vs 9.8%; p-value <0.0001), UST (27.3% vs 16.1%; p-value =0.0422), and VDZ (22.4% vs 11.6%; p-value =0.0069) in the second year post biologic initiation. Proportion of patients with all-cause inpatient stays also decreased in the second year post initiation relative to baseline.
- The mean annual CD-related inpatient costs also decreased significantly by $4,878 for ADA (p-value <0.0001), $7,074 for IFX (p-value <0.0001), and $5,917 for UST (p-value =0.0163) in the second year of follow-up from baseline. A numerical decrease of $698 was also observed for VDZ (p-value =0.6837) during second year follow-up compared to baseline. There were also decreases in the mean annual all-cause inpatient costs for the four biologic cohorts in the second year of follow-up relative to baseline.

Figure 3. Pre-post CD-related and All-cause Emergency Room Visits and Associated Costs Among Bio-naïve Crohn's Disease Patients Initiated Treatment With Biologics

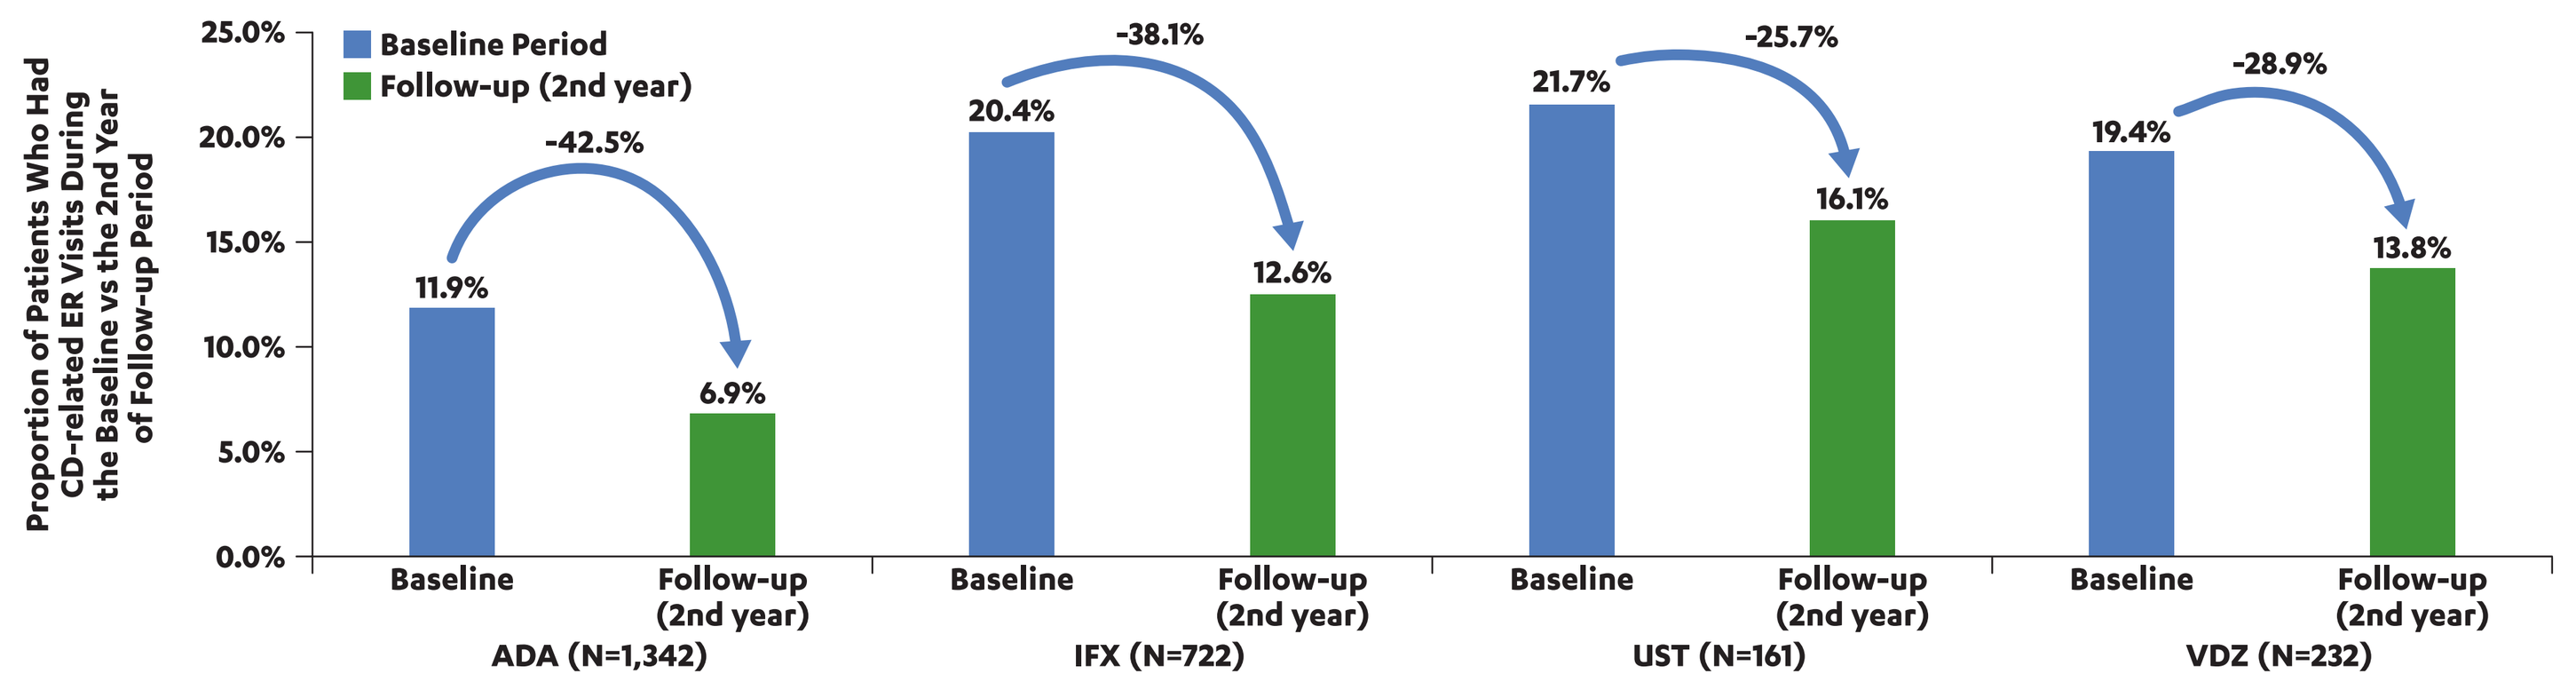
Proportion of Patients Who Had CD-related ER Visits During the Baseline vs the 2nd Year of Follow-up Period
Baseline Period
Follow-up (2nd year)
11.9%
6.9%
-42.5%
20.4%
12.6%
-38.1%
21.7%
16.1%
-25.7%
19.4%
13.8%
-28.9%
Baseline
Follow-up (2nd year)
ADA (N=1,342)
IFX (N=722)
UST (N=161)
VDZ (N=232)

	ADA (N=1,342)		IFX (N=722)		UST (N=161)		VDZ (N=232)	
	Baseline vs Follow-up (2nd year)	Relative change from baseline	Baseline vs Follow-up (2nd year)	Relative change from baseline	Baseline vs Follow-up (2nd year)	Relative change from baseline	Baseline vs Follow-up (2nd year)	Relative change from baseline
CD-related ER utilization	11.9% vs 6.9% (p<0.0001)	-42.5%	20.4% vs 12.6% (p=0.0004)	-38.1%	21.7% vs 16.1% (p=0.3057)	-25.7%	19.4% vs 13.8% (p=0.1715)	-28.9%
Mean annual CD-related ER costs	$340 vs $199 (p=0.0001)	-41.5%	$371 vs $249 (p=0.0558)	-32.8%	$478 vs $373 (p=0.6069)	-22.0%	$425 vs $378 (p=0.7655)	-11.1%
All-cause ER utilization	22.4% vs 13.9% (p<0.0001)	-37.7%	40.7% vs 25.1% (p<0.0001)	-38.4%	41.6% vs 28.6% (p=0.0599)	-31.3%	34.9% vs 28.4% (p=0.2482)	-18.5%
Mean annual all-cause ER costs	$887 vs $530 (p<0.0001)	-40.3%	$873 vs $560 (p=0.0026)	-35.9%	$1,024 vs $850 (p=0.5858)	-17.0%	$865 vs $637 (p=0.2980)	-26.4%

ADA: adalimumab; CD: Crohn's disease; ER: Emergency room; IFX: infliximab; UST: Ustekinumab; VDZ: vedolizumab
Relative change from baseline=(Utilization or cost during follow up-Utilization or cost during baseline)/Utilization or cost during baseline period
Note: Statistical significance represented by p-values (p-value <0.05).

- Proportion of patients with CD-related ER visits decreased significantly for ADA (11.9% vs 6.9%; p-value <0.0001) and IFX (20.4% vs 12.6%; p-value =0.0004) and decreased numerically for UST (21.7% vs 16.1%; p-value =0.3057) and VDZ (19.4% vs 13.8%; p-value =0.1715) in the second year post biologic initiation. Decreases were also observed for all-cause ER visits in the second-year post initiation compared to baseline.
- The mean annual CD-related ER costs showed a decrease of $141 (p-value <0.0001) for ADA, $122 (p-value =0.0558) for IFX, $105 (p-value = 0.6069) for UST, and $47 (p-value =0.7655) for VDZ in the second year of follow-up compared to baseline. Similar trend of decreases was also observed for all-cause ER costs.

Figure 4. Pre-post CD-related Surgeries and Associated Costs Among Bio-naïve Crohn's Disease Patients Initiated Treatment With Biologics

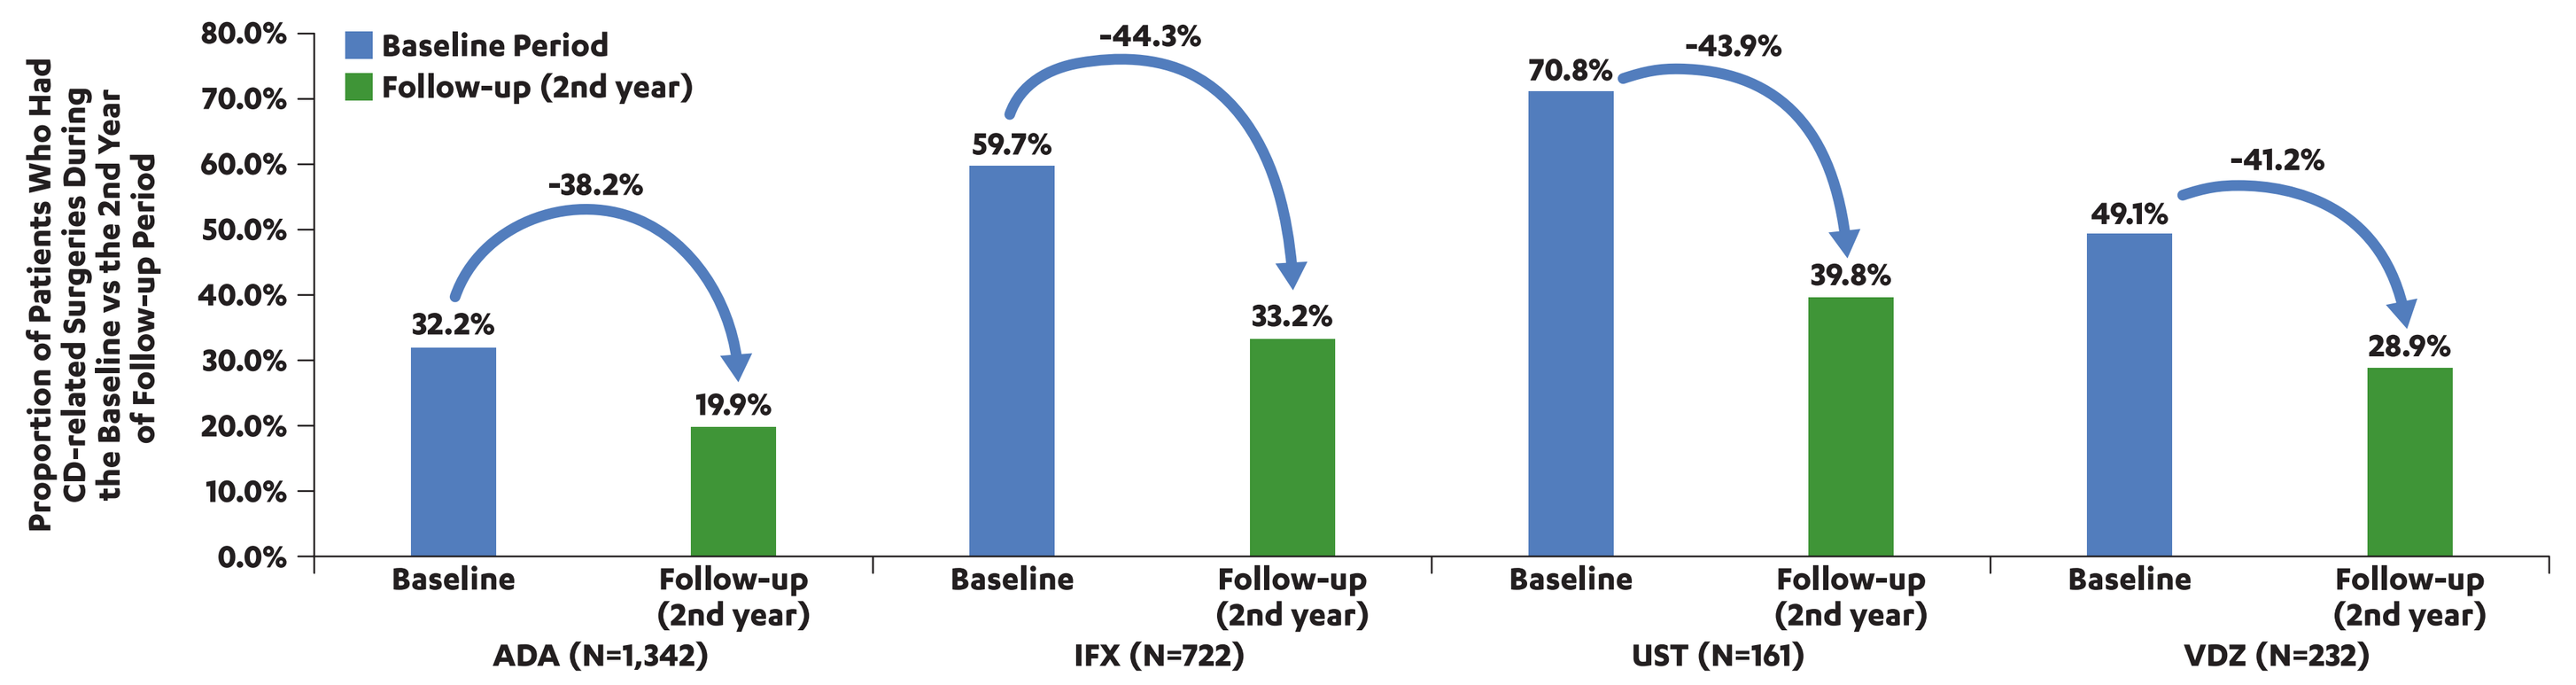
Proportion of Patients Who Had CD-related Surgeries During the Baseline vs the 2nd Year of Follow-up Period
Baseline Period
Follow-up (2nd year)
32.2%
19.9%
-38.2%
59.7%
33.2%
-44.3%
70.8%
39.8%
-43.9%
49.1%
28.9%
-41.2%
Baseline
Follow-up (2nd year)
ADA (N=1,342)
IFX (N=722)
UST (N=161)
VDZ (N=232)

	ADA (N=1,342)		IFX (N=722)		UST (N=161)		VDZ (N=232)	
	Baseline vs Follow-up (2nd year)	Relative change from baseline	Baseline vs Follow-up (2nd year)	Relative change from baseline	Baseline vs Follow-up (2nd year)	Relative change from baseline	Baseline vs Follow-up (2nd year)	Relative change from baseline
CD-related surgeries	32.2% vs 19.9% (p<0.0001)	-38.2%	59.7% vs 33.2% (p<0.0001)	-44.3%	70.8% vs 39.8% (p=0.0002)	-43.9%	49.1% vs 28.9% (p=0.0006)	-41.2%
Mean annual CD-related surgery costs	$2,543 vs $1,153 (p<0.0001)	-54.7%	$3,904 vs $1,728 (p=0.0019)	-55.7%	$3,644 vs $2,091 (p=0.1098)	-42.6%	$2,617 vs $2,214 (p=0.6593)	-15.4%

ADA: adalimumab; CD: Crohn's disease; IFX: infliximab; UST: Ustekinumab; VDZ: vedolizumab
Relative change from baseline=(Utilization or cost during follow up-Utilization or cost during baseline)/Utilization or cost during baseline period
Note: Statistical significance represented by p-values (p-value <0.05).

- Compared to baseline, proportion of patients with CD-related surgeries decreased significantly for ADA (32.2% vs 19.9%; p-value <0.0001), IFX (59.7% vs 33.2%; p-value <0.0001), UST (70.8% vs 39.8%; p-value =0.0002), and VDZ (49.1% vs 28.9%; p-value =0.0006) in the second year post biologic initiation.
- CD-related surgical costs decreased by $1,390 (p-value <0.0001) for ADA, $2,176 (p-value = 0.0019) for IFX, $1,553 (p-value =0.1098) for UST, and $403 (p-value =0.6593) for VDZ during second year post biologic initiation compared to baseline.

LIMITATIONS

- National drug code (NDC) and healthcare common procedure coding system (HCPCS) codes were used to identify all biologic claims. For UST, permanent HCPCS codes for intravenous UST were not available for until January 2018. It is possible that some UST induction doses were not adequately captured.
- Analysis of claims data depends on correct diagnosis, procedure, and drug codes. Coding errors can result in misclassification.
- Patients were designated as bio-naïve based on the absence of a biologic claim in the 1-year baseline period. It is possible that patients may have had biologic use prior to the 1-year baseline period.
- The findings from this study are descriptive in nature, and therefore should not be used to make any comparisons between products studied.

CONCLUSIONS

- There appeared to be varying levels of baseline disease severity among treatments studied.
- Among bio-naïve CD patients that initiated biologics, there were decreases in the proportion of patients with CD-related ER visits, inpatient stays, surgeries, and associated costs from baseline to the second year of follow-up.

REFERENCES

1. Torres, J., Mehandru, S., Colombel, J. F., & Peyrin-Biroulet, L. (2017). Crohn's disease. *The Lancet*, 389(10080), 1741-1755.
2. Baumgart, D. C., & Sandborn, W. J. (2012). Crohn's disease. *The Lancet*, 380(9853), 1590-1605.
3. Floyd DN, Langham S, Séverac HC, Levesque BG. The economic and quality-of-life burden of Crohn's disease in Europe and the United States, 2000 to 2013: A systematic review. *Dig Dis Sci*. 2015;60(2):299-312.
4. Ganz ML, Sugarman R, Wang R, Hansen BB, Håkan-Bloch J. The economic and health-related impact of Crohn's disease in the United States: Evidence from a nationally representative survey. *Inflamm Bowel Dis*. 2016;22(5):1032-1041.
5. Bernstein CN. (2010). Epidemiology of Crohn's disease and ulcerative colitis in a central Canadian province: a population-based study. *Am J Epidemiol*. 1999;149(10):916–24.
6. Obando C. (2020). Persistence, dose titration, and health care resource utilization among Crohn's disease patients treated with ustekinumab: a real-world analysis in the United States. *Adv Ther*. 2020;37(5):2127–43.

DISCLOSURES

Ruizhi Zhao, Zhijie Ding, and Sumesh Kachroo are employees of Janssen Scientific Affairs, LLC and Johnson & Johnson. Anissa Cyhaniuk, and Parul Gupta are employees of SIMR, LLC and supported this study as a paid consultant to Janssen Scientific Affairs.

Presented at the 27th Annual Meeting of the International Society for Pharmacoeconomics and Outcomes Research May 15–18, 2022; National Harbor, Washington, DC

This study was supported by Janssen Scientific Affairs, LLC