Real-World Economic Burden of Illicit-Use Opioid Overdose Patients with and without Respiratory Comorbidity and not on Medication for Opioid Use Disorder

Arun Changolkar, MBA, PhD, MSc[1]; Mohan K. Sindhwani, MD[2]; Shaheen Kaplan, PharmD, MBA[2]; Gay Owens, PharmD, MBA[2]
[1]Emergent BioSolutions, Philadelphia, PA, USA, [2]Emergent BioSolutions, Gaithersburg, MD USA

ISPOR 2022 #ISPORAnnual

Background

Illicit-Use Opioid Overdose in the United States

- 9.5 million people in the United States misused opioids in 2020, including over 900,000 people who used heroin.[1]
- 2.7 million individuals met the criteria for an opioid use disorder in 2020.[1]
 - Medication-assisted treatment helps opioid use disorder patients sustain their recovery and prevent future overdose.[2]
 - 8 in 9 individuals with an opioid use disorder did not receive medication for opioid use disorder in 2020.[1]
- The use of Illicit opioids increases the risk of respiratory depression and overdose death.[3] Furthermore, substantial evidence shows an increased burden of respiratory diseases in illicit opioid users.[4]
- 76,975 people died from an opioid overdose in the 12-month period ending in November 2021, including 67,293 overdose deaths involving synthetic opioids and 9,132 overdose deaths involving heroin.[5]
- Opioid overdoses account for more than $11 billion in annual hospital costs nationwide, according to research by a healthcare improvement company.[6]

Naloxone

- Naloxone is a prescription medication that rapidly counteracts the respiratory depressant effects of opioids and helps prevent death when administered during an opioid overdose.
- The Centers for Disease Control and Prevention recommends prescribing naloxone to all patients at risk for opioid overdose, including illicit opioid users and individuals with a history of overdose or substance use disorder.[3,7]

Objectives

We analyzed a large U.S. healthcare claims database to identify illicit-use opioid overdose patients not on medication for opioid use disorder with respiratory comorbidity (RIOP) and illicit-use opioid overdose patients not on medication for opioid use disorder without respiratory comorbidity (IOP) between January 2016 and December 2019.

Our study assessed:

- Rates of RIOP and IOP by payor (overall, commercial, Medicare, and Medicaid) and state.
- Demographic and clinical characteristics of RIOP and IOP (overall, commercial, Medicare, and Medicaid).
- Proportion of RIOP and IOP administered naloxone at index event (overall, commercial, Medicare, and Medicaid).
- Economic burden associated with treatment of RIOP and IOP (overall, commercial, Medicare, and Medicaid) with and without naloxone treatment at the index event.

Methods

Study Design

- A retrospective cohort study.

Data Source

- Optum's Clinformatics® Data Mart (CDM) that is derived from administrative health claims for members of large Commercial, Medicare Advantage, and Managed Medicaid health plans.[8]

Patient Inclusion Criteria

- ≥18 years old with a diagnosis of an opioid overdose (index event) between January 1, 2016 and December 31, 2019 (index period).
- Minimum continuous health plan enrollment with medical and pharmacy coverage for 365 days both before and after index date (first overdose claim date).
- No prescriptions for an opioid filled in the 365-day pre-index period or on the day of the index event.
- At least one medical claim with a diagnosis of opioid overdose and diagnosis of opioid misuse (opioid abuse, dependence, unspecified opioid use related adverse effects) during the study period.
- No treatment with medication for opioid use disorder (MOUD) prior to the index period.

Analysis

- RIOP and IOP rates were analyzed by insurance coverage (commercial, Medicare, and Medicaid), state, comorbidities, healthcare costs for index event, and total costs with and without naloxone administration at index event. Patient comorbidities were characterized using the Charlson Comorbidities Index (CCI).
- International Classification of Disease (ICD)-9-CM and ICD-10-CM codes (opioid overdose diagnosis) and National Drug Codes were used to search claims in the database and identify RIOP and IOP.

Results

Figure 1. Characteristics of Illicit-Use Opioid Overdose Patients by Payor

	RIOP				IOP			
	Overall n=845	Commercial n=63 (7.5%)	Medicare n=205 (24.3%)	Medicaid n=577 (68.3%)	Overall n=5,633	Commercial n=876 (15.6%)	Medicare n=472 (8.4%)	Medicaid n=4,285 (76.1%)
Age (SD)	52 (16)	35 (16)	68 (11)	48 (14)	40 (15)	30 (12)	66 (13)	39 (13)
Gender								
Female	434 (51.36%)	25 (39.68%)	110 (53.66%)	299 (51.82%)	2,220 (39.41%)	285 (32.53%)	206 (43.64%)	1,729 (40.35%)
Male	411 (48.64%)	38 (60.32%)	95 (46.34%)	278 (48.18%)	3,413 (60.59%)	591 (67.47%)	266 (56.36%)	2,556 (59.65%)
Race								
Caucasian	477 (56.45%)	41 (65.08%)	138 (67.32%)	298 (51.65%)	3,514 (62.38%)	594 (67.81%)	303 (64.19%)	2,617 (61.07%)
African American	153 (18.11%)	7 (11.11%)	31 (15.12%)	115 (19.93%)	842 (14.95%)	53 (6.05%)	69 (14.62%)	720 (16.80%)
Hispanic	33 (3.91%)	8 (12.70%)	10 (4.88%)	15 (2.60%)	255 (4.55%)	95 (10.84%)	33 (6.99%)	127 (2.96%)
Asian	3 (0.36%)	0 (0.00%)	2 (0.98%)	1 (0.17%)	37 (0.66%)	12 (1.37%)	2 (0.42%)	23 (0.54%)
Other	179 (21.18%)	7 (11.11%)	24 (11.71%)	148 (25.65%)	985 (17.49%)	122 (13.93%)	65 (13.77%)	798 (18.62%)
CCI score, mean	2.60	1.94	3.97	2.18	0.77	0.53	2.74	0.60
Patients with at least one other CCI comorbidity	830 (98.22%)	62 (98.41%)	202 (98.54%)	566 (98.09%)	1,878 (33.34%)	243 (27.74%)	350 (74.15%)	1,285 (29.99%)

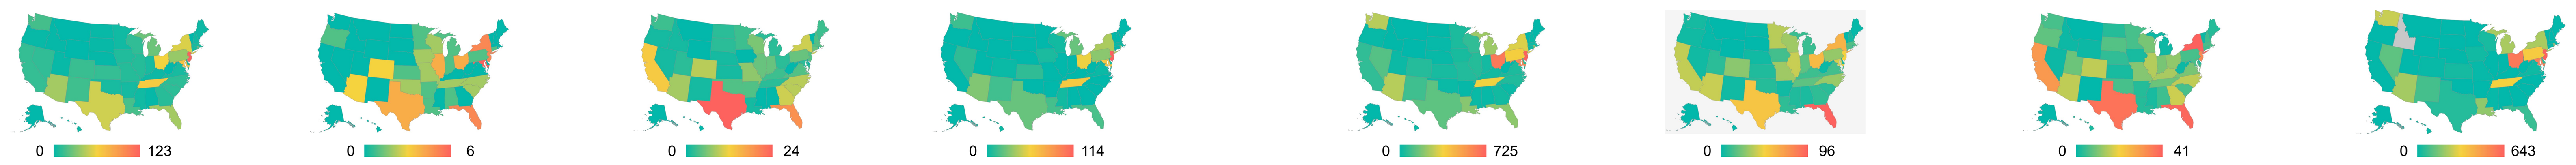
0 123
0 6
0 24
0 114
0 725
0 96
0 41
0 643

Figure 2. Comorbidities Among Prescription Opioid Overdose Patients by Payor

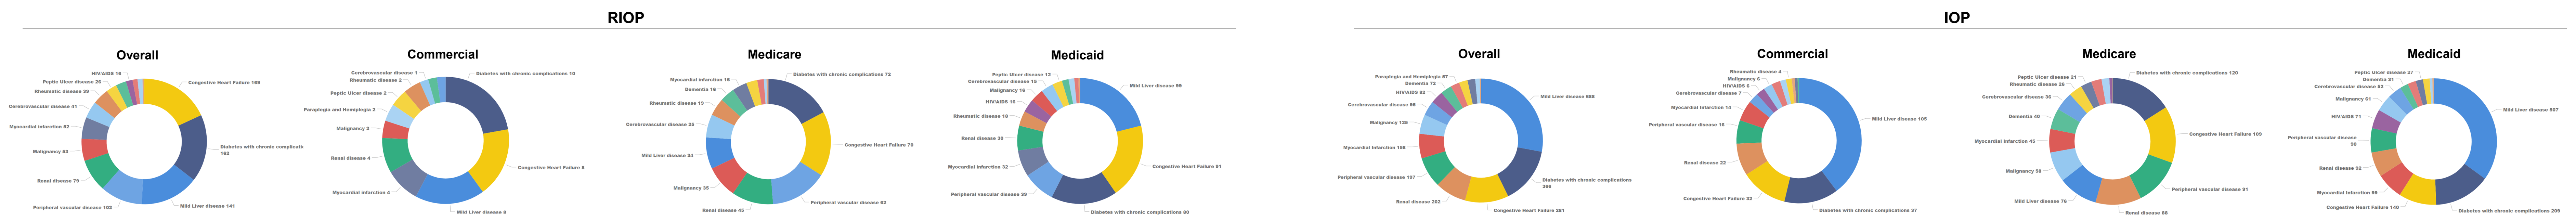
RIOP
Overall
Commercial
Medicare
Medicaid
IOP
Overall
Commercial
Medicare
Medicaid

Figure 3. Administration of Naloxone Among Prescription Opioid Overdose Patients, At-Index

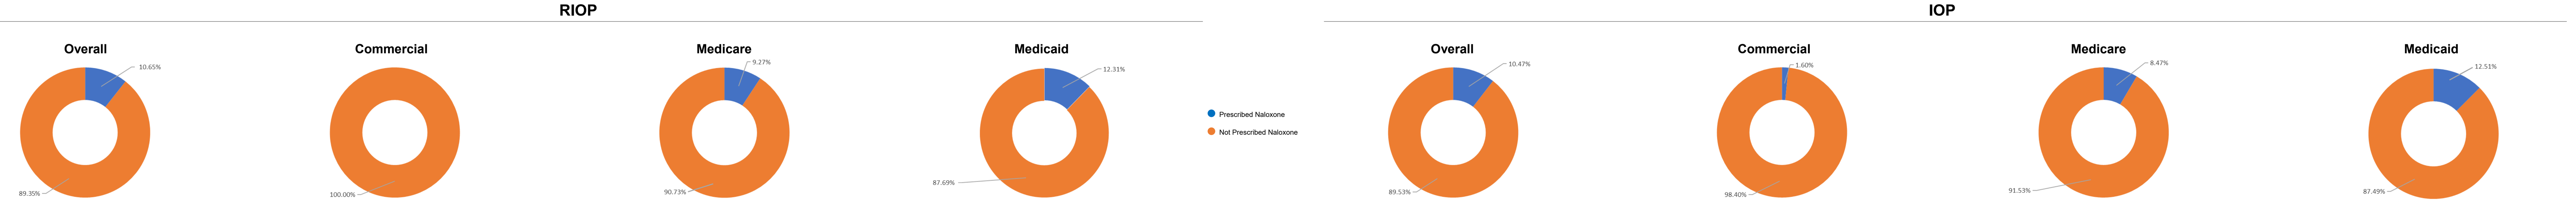
RIOP
Overall
10.65%
89.35%
Commercial
100.00%
Medicare
9.27%
90.73%
Medicaid
12.31%
87.69%
Prescribed Naloxone
Not Prescribed Naloxone
IOP
Overall
10.47%
89.53%
Commercial
1.60%
98.40%
Medicare
8.47%
91.53%
Medicaid
12.51%
87.49%

Figure 4. Inflation-Adjusted Costs Associated with Treatment of Opioid Overdose (At-Index Event)

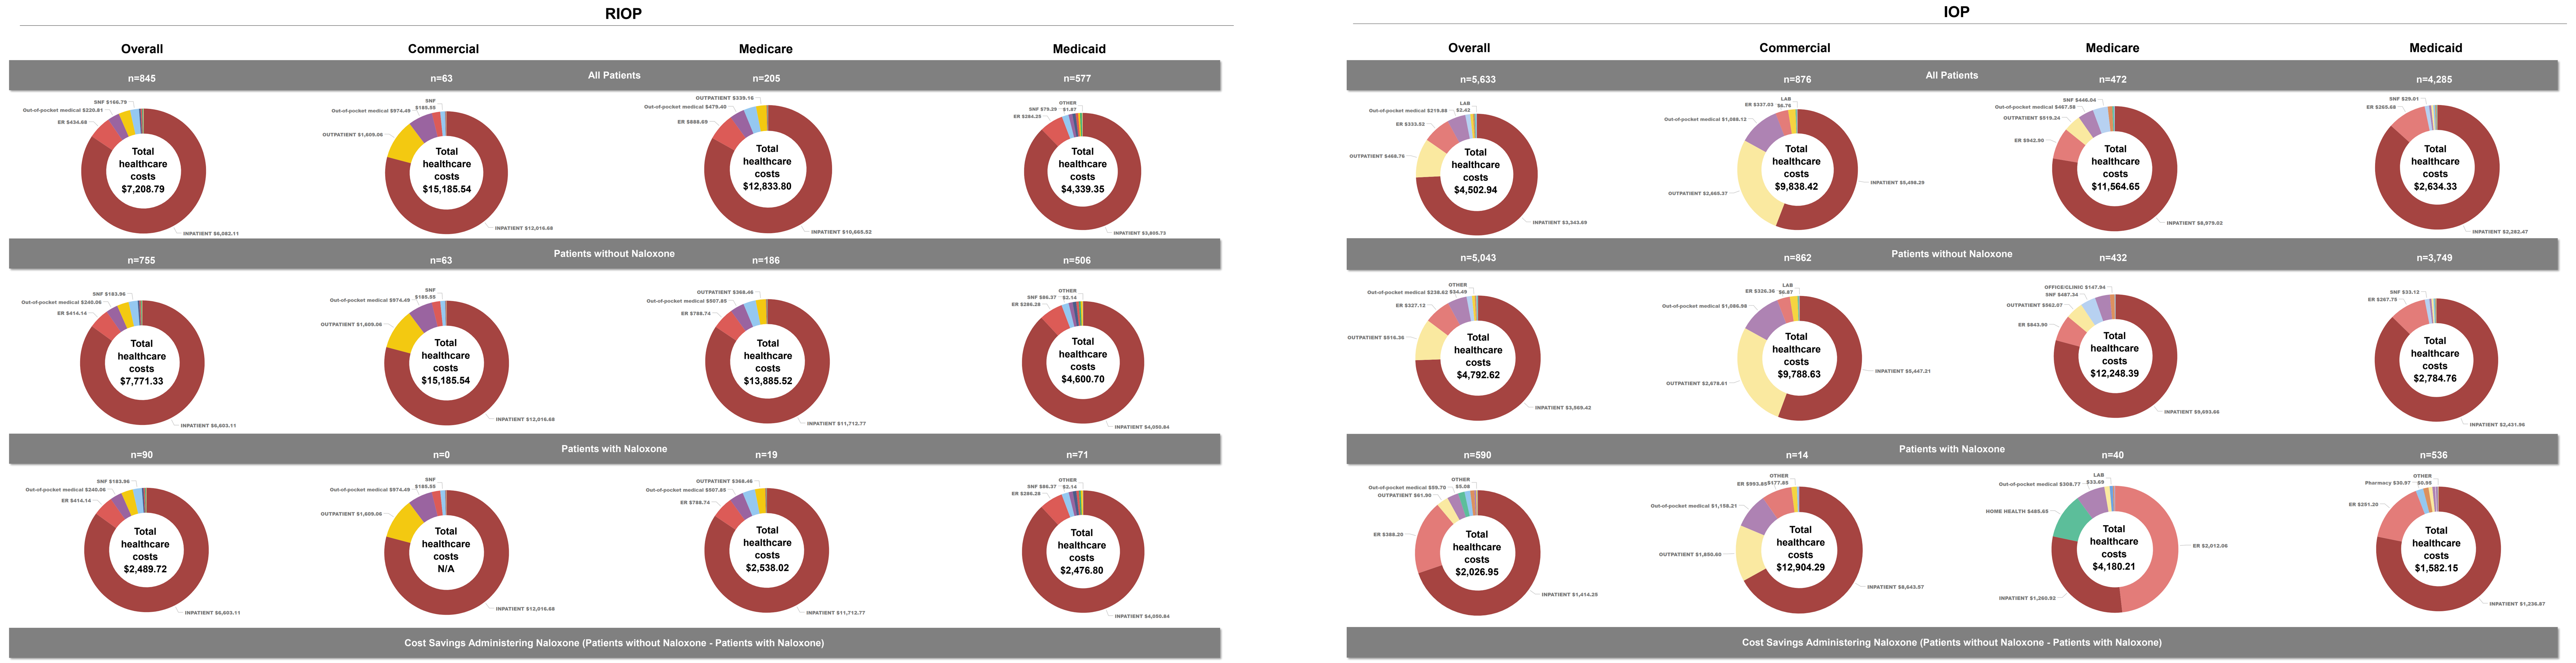
RIOP
Overall
Commercial
Medicare
Medicaid
All Patients
n=845
Total healthcare costs $7,208.79
n=63
Total healthcare costs $15,185.54
n=205
Total healthcare costs $12,833.80
n=577
Total healthcare costs $4,339.35
Patients without Naloxone
n=755
Total healthcare costs $7,771.33
n=63
Total healthcare costs $15,185.54
n=186
Total healthcare costs $13,885.52
n=506
Total healthcare costs $4,600.70
Patients with Naloxone
n=90
Total healthcare costs $2,489.72
n=0
Total healthcare costs N/A
n=19
Total healthcare costs $2,538.02
n=71
Total healthcare costs $2,476.80
IOP
Overall
Commercial
Medicare
Medicaid
All Patients
n=5,633
Total healthcare costs $4,502.94
n=876
Total healthcare costs $9,838.42
n=472
Total healthcare costs $11,564.65
n=4,285
Total healthcare costs $2,634.33
Patients without Naloxone
n=5,043
Total healthcare costs $4,792.62
n=862
Total healthcare costs $9,788.63
n=432
Total healthcare costs $12,248.39
n=3,749
Total healthcare costs $2,784.76
Patients with Naloxone
n=590
Total healthcare costs $2,026.95
n=14
Total healthcare costs $12,904.29
n=40
Total healthcare costs $4,180.21
n=536
Total healthcare costs $1,582.15

Cost Savings Administering Naloxone (Patients without Naloxone - Patients with Naloxone)

	RIOP Overall	RIOP Commercial	RIOP Medicare	RIOP Medicaid	IOP Overall	IOP Commercial	IOP Medicare	IOP Medicaid
Cost savings	$5,281.61	Cannot be determined	$11,347.50	$2,123.90	$2,765.67	-$3,115.66	$8,068.18	$1,202.61

Key Findings

Patient Characteristics

- 6,478 illicit-use opioid overdose patients not on medication for opioid use disorder were identified between 2016 and 2019. This included 845 illicit-use opioid overdose patients not on medication for opioid use disorder with respiratory comorbidity (RIOP) and 5,633 illicit-use opioid overdose patients not on medication for opioid use disorder without respiratory comorbidity (IOP).
- The average age of the RIOP cohort was 52 years (SD±16), with the highest average age among the Medicare population (68) followed by those with Medicaid (48) and commercial insurance (35). RIOP were mostly female (51.36%), Caucasian (56.45%), and covered by Medicaid (68.28%). States with the highest RIOP rates were NJ (14.56%), MD (8.28%), TN (7.81%), OH (7.22%), and NY (6.51%).
- The average age of the IOP cohort was 40 years (SD±15), with the highest average age among the Medicare population (66) followed by those with Medicaid (39) and commercial insurance (30). The majority of IOP were male (60.59%), Caucasian (62.38%), and insured by Medicaid (76.07%). States with the highest number of HDOOP included NJ (12.87%), OH (11.54%), MD (10.74%), PA (6.73%), and TN (6.30%).

Comorbidities among RIOP and IOP

- One-third (33.34%) of IOP had at least one comorbidity and nearly all (98.22%) RIOP had at least one other comorbidity in addition to a respiratory comorbidity.
- Average Charlson Comorbidity Index (CCI) scores were higher among RIOP (2.60) than IOP (0.77). Medicare patients were the sickest individuals among both RIOP (3.97) and IOP (2.74). In the RIOP cohort, the Medicaid (2.18) and commercial insurance (1.94) groups had similar average CCI scores. This trend was also observed among the Medicaid (0.60) commercial insurance (0.53) groups within the IOP cohort.
- The most common CCI comorbidities (other than respiratory comorbidities) among the RIOP cohort for all three payor groups included: diabetes with chronic complications (14.13%-35.64%), congestive heart failure (12.90%-34.65%), mild liver disease (12.90%-16.83%), myocardial infarction (6.45%-7.92%), renal disease (5.30%-22.28%), malignancy (3.23%-17.33%), and peripheral vascular disease (1.61%-30.69%). The most common CCI comorbidities among the IOP cohort included: mild liver disease (21.71%-43.21%), diabetes with chronic complications (15.23%-34.29%), congestive heart failure (10.89%-31.14%), peripheral vascular disease (7.00%-26.00%), renal disease (7.16%-25.14%), and myocardial infarction (5.76%-12.86%).

Naloxone Use and Healthcare Utilization

- Only 10.65% of RIOP received naloxone at index event. Medicaid patients received naloxone at the highest rate (12.31%), followed by those with Medicare (9.27%). None of the RIOP with commercial insurance received naloxone. In the IOP cohort, 10.47% of patients received naloxone at index event, with Medicaid (12.51%) and Medicare (8.47%) patients receiving naloxone at a much higher rate than commercial patients (1.60%).
- Among RIOP who received naloxone at index event, 99.89% needed emergency care, 67.78% received inpatient treatment, and 10.00% required advanced medical care. Among those in the RIOP cohort not administered naloxone, 87.95% needed emergency care, 47.42% received inpatient treatment, and 28.34% required advanced medical care.
- Among IOP who were provided naloxone, 98.64% needed emergency care, 17.12% were admitted for inpatient treatment, and 6.61% required advanced medical care. Among those in the IOP cohort not treated with naloxone, 90.68% needed emergency care, 29.31% received inpatient treatment, and 14.55% required advanced medical care.
- Average index event costs were higher among RIOP ($7,208.79) than IOP ($4,502.94), and costs were noticeably different among the 3 payors. Total costs for RIOP with commercial insurance ($15,185.54) and Medicare ($12,833.80) were much higher than for those with Medicaid ($4,339.95), while a similar trend was also observed among IOP with Medicare ($11,564.65) and commercial groups ($9,838.42) showing much higher average costs than the Medicaid ($2,634.33) cohort.
- Total costs for RIOP not treated with naloxone were $7,771.33, more than triple the total costs for RIOP treated with naloxone ($2,489.72). IOP not administered naloxone had total costs of $4,792.62, more than double the costs of IOP provided with naloxone at index event ($2,026.95).
- Based on the total medical cost differences between Medicare patients treated with naloxone ($2,538.02) and Medicare patients not treated with naloxone ($13,885.52), we estimate that 100,000 RIOP provided with naloxone at the index event will result in ~$1.13 billion savings to Medicare plans. Additionally, at-index naloxone treatment for 100,000 RIOP would amount to ~$212 million in savings for Medicaid. Cost savings for commercial insurance payors cannot be assessed because no RIOP with commercial insurance coverage received naloxone at the index event (n=0).
- A conservative estimate of 100,000 IOP index events shows that naloxone treatment can yield cost savings of ~$807 million for Medicare and ~$120 million for Medicaid. Commercial insurance cost savings cannot be assessed due to the low number of commercial insurance beneficiaries (n=14) that received naloxone in the IOP cohort.

Limitations

- Inherent limitations of claims data analysis include the possibility of incomplete, inaccurate, or missing data. Undetected coding errors could have resulted in the misidentification of RIOP and IOP. Moreover, our use of claims data only allowed for analysis of dispensed prescriptions and diagnoses if a claim was filed. For example, opioid overdose events and opioid use disorder medication dispensing that occurred outside of medical channels are not captured in the data used in this study.
- This study presents illicit-use opioid overdose rates only. Although we limited our cohort population to opioid overdose patients without an opioid prescription, we cannot rule out the possibility that some of our observed opioid overdoses were attributed to the use of prescription opioids that were dispensed to the patient prior to the pre-index period.
- Due to the continuous enrollment constraint of our study, our findings did not include overdose-induced deaths occurring at the index event.
- Calculated at-index costs were conservative because they do not account for any subsequent overdose events and associated medical encounters for treatment occurring within the same year.
- One needs to use caution and consider the implications of smaller sample size in some of the groups while interpreting these study results

Conclusions

- Our study provides previously unavailable information on RIOP and IOP rates broken down by payor types (overall, commercial, Medicare, and Medicaid), patient characteristics, and comorbidities.
- RIOP and IOP demonstrated significant economic burden to the U.S. healthcare system and payors of commercial, Medicare, and Medicaid plans.
- Overall, the RIOP cohort had higher average index event total costs than the IOP group. These cost differences are likely due to the higher rates of underlying comorbidities among RIOP resulting in the need for more costly services such as inpatient treatment and advanced medical care.
- Although naloxone use greatly reduced medical costs and healthcare utilization following an illicit-use opioid overdose, this medication remains grossly under-dispensed among RIOP and IOP.
- Given increasingly constrained healthcare budgets, higher rates of naloxone co-prescribing may help reduce the economic burden associated with treating RIOP and IOP.
- Our findings may serve as a useful resource for policymakers, researchers, and payors to further evaluate the real-world benefits of naloxone among RIOP and IOP populations.

Future Research

- Further studies are needed to explore the real-world value of naloxone prescribing among RIOP and IOP populations.
- Future research should explore the relationship between naloxone use, lower medical costs, and decreased utilization of healthcare resources among RIOP and IOP.
- Additional studies are needed to assess the benefits of naloxone prescribing among RIOP and IOP using data from states that have implemented co-prescribing policies.

Acknowledgments

The authors of this presentation would like to thank ScioScientific and the Power BI team from Emergent BioSolutions for their contributions to this poster.

References

1. Substance Abuse and Mental Health Services Administration. (2021). *Key substance use and mental health indicators in the United States: Results from the 2020 National Survey on Drug Use and Health* (HHS Publication No. PEP21-07-01-003, NSDUH Series H-56). Rockville, MD: Center for Behavioral Health Statistics and Quality, Substance Abuse and Mental Health Services Administration.
2. Substance Abuse and Mental Health Services Administration. Medication-Assisted Treatment (MAT). https://www.samhsa.gov/medication-assisted-treatment;retrieved March 26, 2022.
3. Dowell D, Haegerich TM, Chou R. CDC Guideline for Prescribing Opioids for Chronic Pain — United States, 2016. MMWR *Recomm* Rep. 2016;65(No. RR-1):1–49.
4. Hulin J, Brodie A, Stevens J, Mitchell C. Prevalence of respiratory conditions among people who use illicit opioids: a systematic review. *Addiction*. 2020 May;115(5):832-849.
5. National Center for Health Statistics. Provisional Drug Overdose Death Counts. https://www.cdc.gov/nchs/nvss/vsrr/drug-overdose-data.htm; retrieved April 20, 2022.
6. Premier Inc. "Opioid Overdoses Costing U.S. Hospitals an Estimated $11 Billion Annually," news release, Jan. 3, 2019. https://www.premierinc.com/newsroom/press-releases/opioid-overdoses-costing-u-s-hospitals-an-estimated-11-billion-annually; retrieved February 8, 2022.
7. Centers for Disease Control and Prevention. Life-Saving Naloxone from Pharmacies. https://www.cdc.gov/vitalsigns/naloxone/index.html; retrieved March 9, 2022.
8. Optum's de-identified Clinformatics® Data Mart Database (2007-2021).