EMERGENT

Real-World Economic Benefit of Naloxone Use Among Illicit-Use Opioid Overdose Patients with Respiratory Comorbidity and on Medication for Opioid Use Disorder (MOUD)

Arun Changolkar, MBA, PhD, MSc[1]; Mohan K. Sindhwani, MD[2]; Shaheen Kaplan, PharmD, MBA[2]; Gay Owens, PharmD, MBA[2]

[1]Emergent BioSolutions, Philadelphia, PA USA, [2]Emergent BioSolutions, Gaithersburg, MD USA

ISPOR2022 #ISPORAnnual

Background

Illicit-Use Opioid Overdose in the United States

- 9.5 million people in the United States misused opioids in 2020.[1]
- 2.7 million individuals met the criteria for an opioid use disorder (OUD) in 2020.[1]
- Nearly 550,000 Americans died from an opioid overdose from 2000 to 2020.[2]
- 68,630 people died from an opioid overdose in 2020, including 13,165 overdose deaths involving heroin, 16,416 overdose deaths involving prescription opioids, and 56,516 overdose deaths involving illicit synthetic opioids (primarily fentanyl).[2]
- Opioid overdoses account for more than $11 billion in annual hospital costs nationwide.[3]

Respiratory Comorbidity among Illicit Opioid Users

- People who use illicit opioids exhibit an increased burden of respiratory diseases.[4-5]
- Fentanyl use among individuals with severe respiratory disease significantly elevates risk for overdose mortality.[6]

Medication for Opioid Use Disorder

- 1 in 9 individuals with an opioid use disorder received medication for opioid use disorder (MOUD) in 2020.[1]
- Some patients on MOUD are at increased risk for overdose due to the loss of tolerance to opioids during treatment.[7]
- Undetected respiratory depression is common in OUD patients receiving MOUD.[8]

Naloxone

- Naloxone is a life-saving medication that blocks the effects of opioids. When administered during an opioid emergency, naloxone can quickly reverse an overdose and help restore normal breathing.
- Current prescription guidelines recommend prescribing naloxone to patients at risk for opioid overdose, even if the patient does not have a current opioid prescription.[9]
- Increased distribution of naloxone can help reduce opioid overdose deaths.[10-11]

Objectives

We analyzed a large U.S. healthcare claims database to identify illicit-use opioid overdose patients with respiratory comorbidity on medication for opioid use disorder (RIOP-RX) and illicit-use opioid overdose patients without respiratory comorbidity on medication for opioid use disorder (IOP-RX) from January 2016 through December 2019.

Our study assessed:

- Rates of RIOP-RX and IOP-RX by payor (overall, commercial, Medicare, and Medicaid) and state.
- RIOP-RX and IOP-RX demographic and clinical characteristics (overall, Commercial, Medicare, and Medicaid).
- Proportion of RIOP-RX and IOP-RX administered naloxone at index event (overall, Commercial, Medicare, and Medicaid).
- Economic burden of RIOP-RX and IOP-RX (overall, commercial, Medicare, and Medicaid), as well as with and without naloxone treatment at the index event.

Methods

Study Design and Data Source

- A retrospective cohort study.

Data Source

- Optum's Clinformatics® Data Mart (CDM) that is derived from administrative health claims for members of large Commercial, Medicare Advantage, and Managed Medicaid health plans.[12]

Patient Cohort

- ≥18 years old with a diagnosis of an opioid overdose (index event = first OOD claim date) between January 1, 2016 and December 31, 2019 (index period).
- Minimum continuous health plan enrollment with medical and pharmacy coverage for 365 days before index date (baseline period) and 365 days after index date (follow-up period).
- No prescriptions for an opioid filled in the 365-day pre-index period or on the day of the index event.
- At least one medical claim with a diagnosis of opioid overdose and diagnosis of opioid misuse (opioid abuse, dependence, unspecified opioid use related adverse effects) during the study period.
- Treatment with medication for opioid use disorder (MOUD) prior to the index period.

Analysis

- RIOP-RX and IOP-RX rates were analyzed by insurance coverage (commercial, Medicare, and Medicare), state, comorbidities, healthcare costs for index event, and total costs with and without naloxone administration at index event. Patient comorbidities were characterized using the Charlson Comorbidities Index (CCI).
- International Classification of Disease (ICD)-9-CM and ICD-10-CM codes (opioid overdose diagnosis) and National Drug Codes (MOUD) were used to search claims in the database and identify RIOP-RX and IOP-RX.

Results

Figure 1. Characteristics of Illicit-Use Opioid Overdose Patients on Medication for Opioid Use Disorder (MOUD), by Payor

	Illicit-Use Opioid Overdose Patients on MOUD without Respiratory Comorbidity (IOP-RX)				Illicit-Use Opioid Overdose Patients on MOUD with Respiratory Comorbidity (RIOP-RX)			
	Overall n=1,494	Commercial n=270 (18.07%)	Medicare n=63 (4.22%)	Medicaid n=1,161 (77.71%)	Overall n=176	Commercial n=22 (12.50%)	Medicare n=19 (10.80%)	Medicaid n=135 (76.70%)
Age								
18-29	548 (36.68%)	207 (76.67%)	0 (0.00%)	341 (29.37%)	38 (21.59%)	13 (59.09%)	0 (0.00%)	25 (18.52%)
30-49	768 (51.41%)	54 (20.00%)	16 (25.40%)	698 (60.12%)	74 (42.05%)	5 (22.73%)	3 (15.79%)	66 (48.89%)
50-64	156 (10.44%)	8 (2.96%)	36 (57.14%)	112 (9.65%)	54 (30.68%)	4 (18.18%)	10 (52.63%)	40 (29.63%)
65-84	22 (1.47%)	1 (0.37%)	11 (17.64%)	10 (0.86%)	9 (5.11%)	0 (0.00%)	6 (31.58%)	3 (2.22%)
≥85	0 (0.00%)	0 (0.00%)	0 (0.00%)	0 (0.00%)	1 (0.57%)	0 (0.00%)	0 (0.00%)	1 (0.74%)
Age, mean (SD)	35 (11)	27 (8)	55 (12)	36 (10)	43 (14)	32 (13)	57 (10)	43 (13)
Gender								
Female	550 (36.81%)	80 (29.63%)	15 (23.81%)	455 (39.19%)	84 (47.73%)	6 (27.27%)	6 (31.58%)	72 (53.33%)
Male	944 (63.19%)	190 (70.37%)	48 (76.19%)	706 (60.81%)	92 (52.27%)	16 (72.73%)	13 (68.42%)	63 (46.67%)
Race								
Caucasian	1,103 (73.83%)	197 (72.96%)	35 (55.56%)	871 (75.02%)	103 (58.52%)	16 (72.73%)	11 (57.89%)	76 (56.30%)
African American	114 (7.63%)	8 (2.96%)	13 (20.63%)	93 (8.01%)	26 (14.77%)	2 (9.09%)	2 (10.53%)	22 (16.30%)
Hispanic	55 (3.68%)	16 (5.93%)	10 (15.87%)	29 (2.50%)	5 (2.84%)	0 (0.00%)	2 (10.53%)	3 (2.22%)
Asian	9 (0.60%)	6 (2.22%)	0 (0.00%)	3 (0.26%)	2 (1.14%)	0 (0.00%)	1 (5.26%)	1 (0.74%)
Other	213 (14.26%)	43 (15.93%)	5 (7.94%)	165 (14.21%)	40 (22.73%)	4 (18.2%)	3 (15.79%)	33 (22.44%)
CCI score, mean	0.58	0.48	1.97	0.53	2.27	1.27	4.58	2.11
Patients with at least one other CCI comorbidity	496 (33.20%)	85 (31.48%)	47 (74.60%)	364 (31.35%)	168 (95.45%)	17 (77.27%)	19 (100.00%)	132 (97.78%)

Illicit opioid overdose rates by state

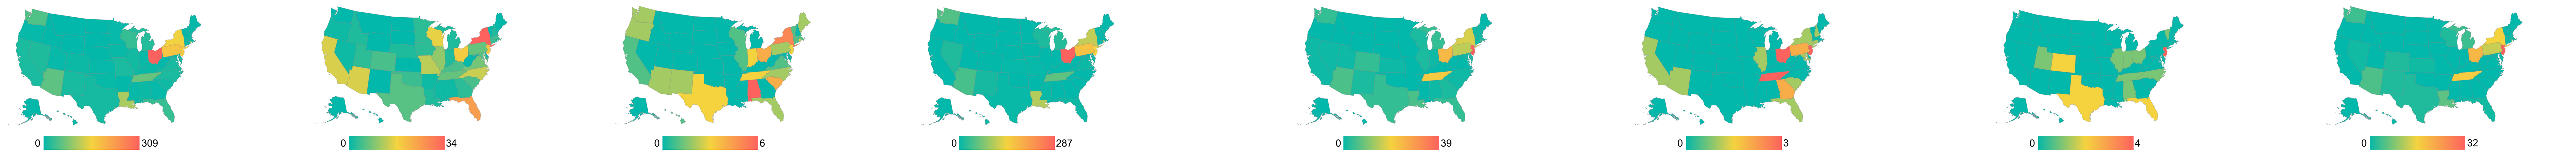

0 309
0 34
0 6
0 287
0 39
0 3
0 4
0 32

Figure 2. Comorbidities Among Illicit-Use Opioid Overdose Patients on MOUD, by Payor

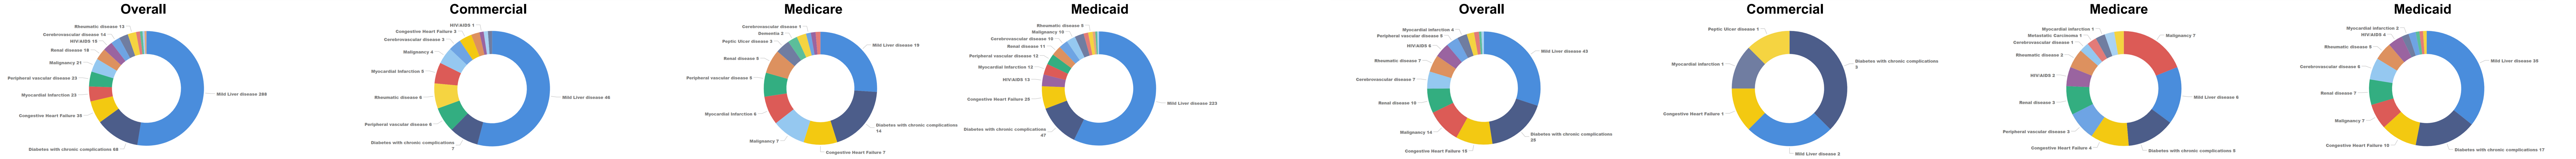

IOP-RX
Overall
Commercial
Medicare
Medicaid
RIOP-RX
Overall
Commercial
Medicare
Medicaid
Mild Liver disease 288
Diabetes with chronic complications 68
Congestive Heart Failure 35
Myocardial Infarction 23
Peripheral vascular disease 23
Malignancy 21
Renal disease 18
HIV/AIDS 15
Cerebrovascular disease 14
Rheumatic disease 13
Mild Liver disease 46
Mild Liver disease 19
Mild Liver disease 223
Mild Liver disease 43
Mild Liver disease 2
Mild Liver disease 6
Mild Liver disease 35

Figure 3. Administration of Naloxone Among Illicit-Use Opioid Overdose Patients on MOUD, At-Index

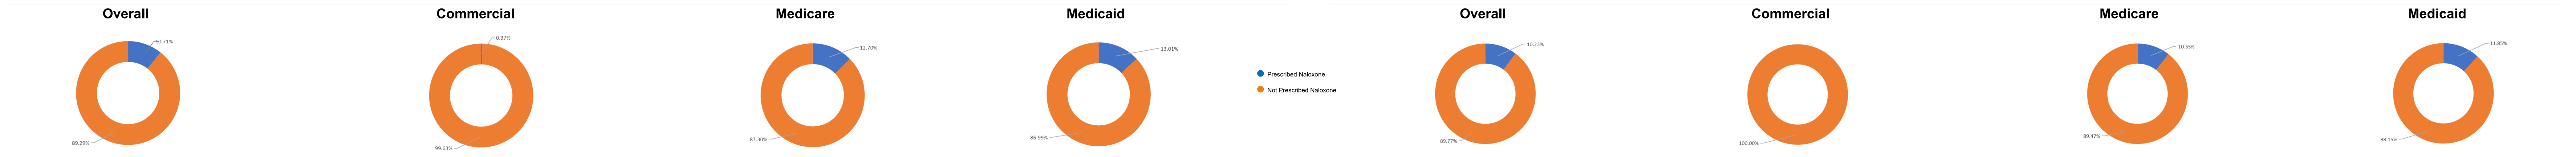

IOP-RX
Overall
Commercial
Medicare
Medicaid
10.71%
89.29%
0.37%
99.63%
12.70%
87.30%
13.01%
86.99%
Prescribed Naloxone
Not Prescribed Naloxone
RIOP-RX
Overall
Commercial
Medicare
Medicaid
10.23%
89.77%
100.00%
10.53%
89.47%
11.85%
88.15%

Figure 4. Inflation-Adjusted Costs Associated with Treatment of Opioid Overdose (At-Index Event)

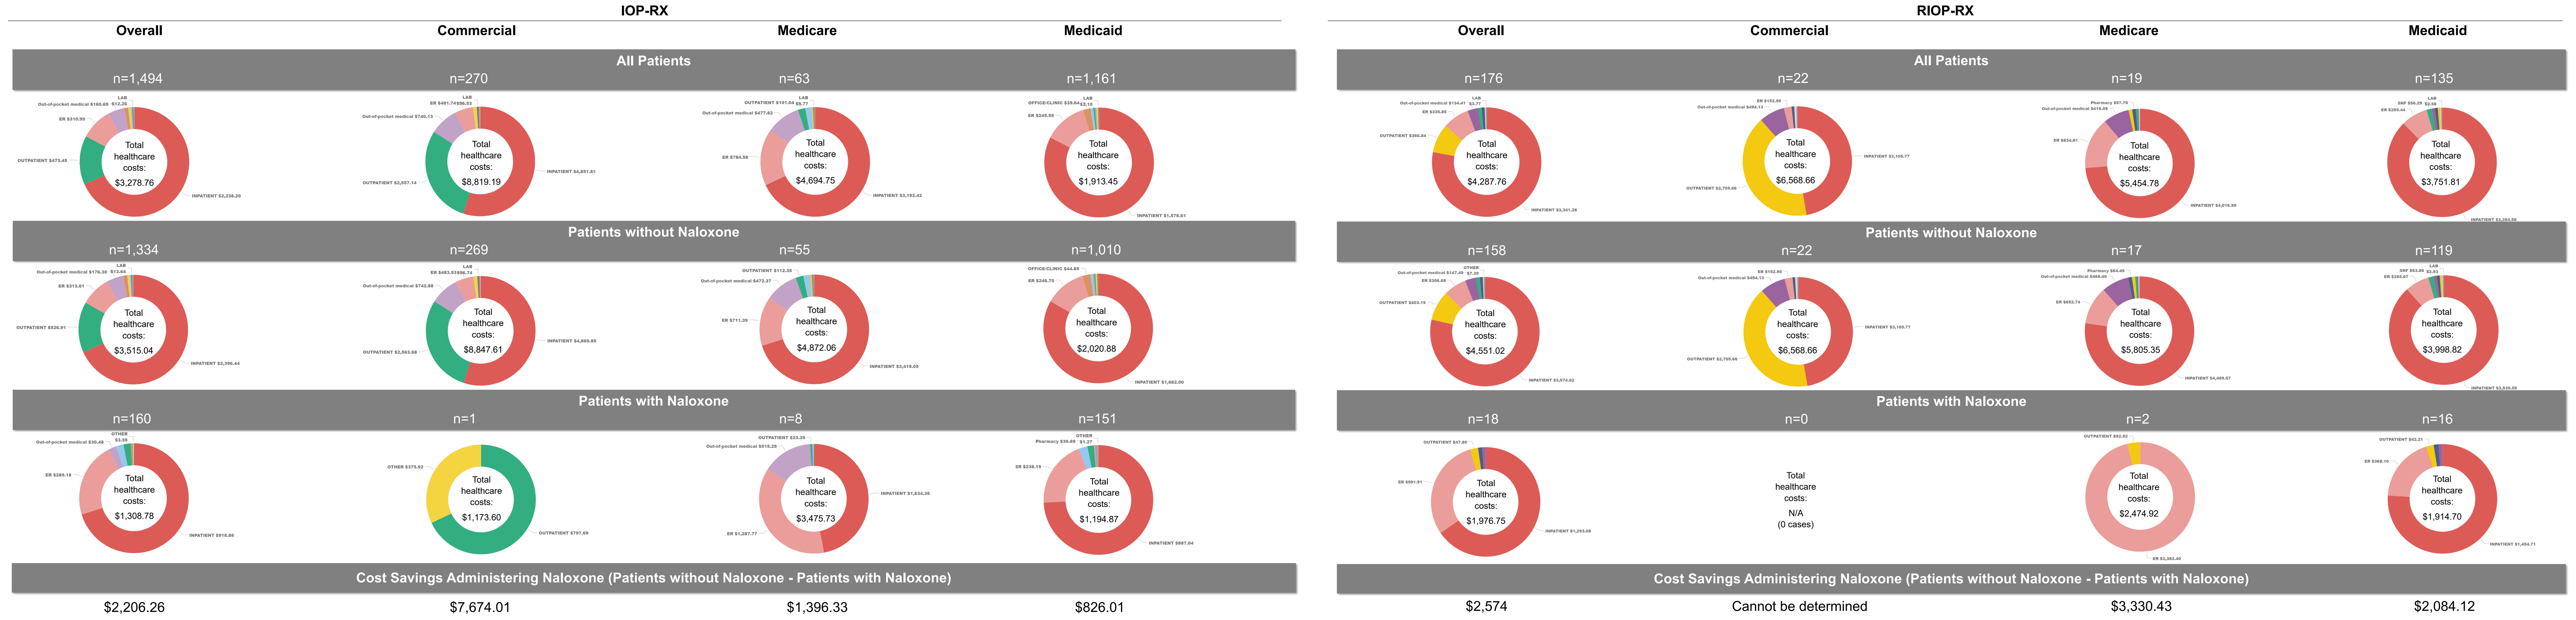

IOP-RX
Overall
Commercial
Medicare
Medicaid
RIOP-RX
Overall
Commercial
Medicare
Medicaid
All Patients
n=1,494
n=270
n=63
n=1,161
n=176
n=22
n=19
n=135
Total healthcare costs: $3,278.76
Total healthcare costs: $8,819.19
Total healthcare costs: $4,694.75
Total healthcare costs: $1,913.45
Total healthcare costs: $4,287.76
Total healthcare costs: $6,568.66
Total healthcare costs: $5,454.78
Total healthcare costs: $3,751.81
Patients without Naloxone
n=1,334
n=269
n=55
n=1,010
n=158
n=22
n=17
n=119
Total healthcare costs: $3,515.04
Total healthcare costs: $8,847.61
Total healthcare costs: $4,872.06
Total healthcare costs: $2,020.88
Total healthcare costs: $4,551.02
Total healthcare costs: $6,568.66
Total healthcare costs: $5,805.35
Total healthcare costs: $3,998.82
Patients with Naloxone
n=160
n=1
n=8
n=151
n=18
n=0
n=2
n=16
Total healthcare costs: $1,308.78
Total healthcare costs: $1,173.60
Total healthcare costs: $3,475.73
Total healthcare costs: $1,194.87
Total healthcare costs: $1,976.75
Total healthcare costs: N/A (0 cases)
Total healthcare costs: $2,474.92
Total healthcare costs: $1,914.70
Cost Savings Administering Naloxone (Patients without Naloxone - Patients with Naloxone)
$2,206.26
$7,674.01
$1,396.33
$826.01
Cost Savings Administering Naloxone (Patients without Naloxone - Patients with Naloxone)
$2,574
Cannot be determined
$3,330.43
$2,084.12

Key Findings

Patient Characteristics

- From 2016-2019, we identified 176 illicit-use opioid overdose patients with respiratory comorbidity who were on medication for opioid use disorder (RIOP-RX) and 1,494 illicit-use opioid overdose patients without respiratory comorbidity who were on medication for opioid use disorder (IOP-RX).
- The average age of the RIOP-RX cohort was 43 years (SD±14), with the highest average age among the Medicare population (57) followed by those with Medicaid (43) and commercial insurance (32). RIOP-RX were mostly male (52.27%), Caucasian (58.52%), and covered by Medicaid (76.70%). Overall, 36.36% of RIOP-RX patients were ≥ 50 years old.
- The average age of the IOP-RX cohort was 35 years (SD±11), with the highest average age among the Medicare population (55) followed by those with Medicaid (36) and commercial insurance (27). The majority of IOP-RX were male (63.19%), Caucasian (78.83%), and insured by Medicaid (77.71%). Overall, 11.91% of IOP-RX patients were ≥ 50 years old.

Rates by State and Payer

- States with the overall highest RIOP-RX rates were NJ (22.16%), OH (13.64%), TN (11.93%), NY (9.66%), and PA (8.52%).
- States with the overall highest number of IOP-RX included OH (20.68%), PA (11.65%), NJ (11.18%), NY (10.17%), and LA (7.16%).

Comorbidities among RIOP-RX and IOP-RX

- One-third (33.20%) of IOP-RX had at least one comorbidity and nearly all (95.45%) RIOP-RX had at least one other comorbidity in addition to a respiratory comorbidity.
- Average Charlson Comorbidity Index (CCI) scores were higher among RIOP-RX (2.27) than IOP-RX (0.58). Medicare patients were the sickest individuals among both RIOP-RX (4.58) and IOP-RX (1.97). Medicaid (0.53) and commercial insurance (0.48) patients within the IOP-RX cohort had similar low CCI scores, while among RIOP-RX the average CCI score for Medicaid (2.11) patients was slightly higher than for those with commercial insurance (1.27).
- The most common CCI comorbidities among the IOP-RX cohort included mild liver disease (40.43%-61.26%), diabetes with chronic complications (8.24%-29.79%), congestive heart failure (3.53%-14.89%), malignancy (2.75%-14.89%), myocardial infarction (3.30%-12.77%), peripheral vascular disease (3.30%-10.64%), rheumatic disease (1.37%-7.06%), and HIV-AIDS (1%-4%). The most common CCI comorbidities (other than respiratory comorbidities) among the RIOP-RX cohort for all three payor groups included mild liver disease (11.76%-31.58%), diabetes with chronic complications (12.88%-26.32%), congestive heart failure (5.88%-21.05%), malignancy (0.00%-36.84%), renal disease (0%-15.79%), peripheral vascular disease (0.00%-15.79%), myocardial infarction (1.52%-5.88%), and peptic ulcer disease (0.76%-5.88%).

Naloxone Use and Healthcare Utilization

- Only 10.23% of RIOP-RX received naloxone at index event. Medicaid patients received naloxone at the highest rate (11.85%), followed by those with Medicare (10.53%). No RIOP-RX with commercial insurance received naloxone. In the IOP-RX cohort, 10.71% of patients received naloxone at index event, with Medicaid (13.01%) and Medicare (12.70%) patients receiving naloxone at a much higher rate than commercial patients (0.37%).
- Among RIOP-RX who received naloxone at index event, 100.00% needed emergency care, 27.78% received inpatient treatment, and 5.56% required advanced medical care. Among those in the RIOP-RX cohort not administered naloxone, 86.08% needed emergency care, 34.81% received inpatient treatment, and 20.25% required advanced medical care.
- Among IOP-RX who were provided naloxone, 98.13% needed emergency care, 8.75% were admitted for inpatient treatment, and 1.25% required advanced medical care. Among those in the IOP-RX cohort not treated with naloxone, 91.25% needed emergency care, 20.99% received inpatient treatment, and 9.37% required advanced medical care.
- Average index event costs were higher among RIOP-RX ($4,287.76) than IOP-RX ($3,278.76), and costs were noticeably different among the 3 payors. In the RIOP-RX cohort, total costs for patients with commercial insurance ($6,568.66) and Medicare ($5,454.78) were comparable and much higher than for those with Medicaid ($3,751.81). In the IOP-RX cohort, total costs for the commercial group ($8,819.19) were nearly double the Medicare group ($4,694.75) and more than four-fold higher than the Medicaid group ($1,913.45).
- Total costs for RIOP-RX not treated with naloxone were $4,551.02, more than double the costs for RIOP-RX treated with naloxone ($1,976.95). IOP-RX not administered naloxone had total costs of $3,515.04, nearly triple the costs of IOP-RX provided with naloxone at index event ($1,308.78).
- Based on the total medical costs differences between patients treated with and without naloxone, we estimate that 100,000 RIOP-RX provided naloxone at index event will result in ~$333 million savings to managed Medicare plans and ~$208 million in savings for managed Medicaid. Cost savings for commercial insurance payors cannot be assessed because no RIOP-RX with commercial insurance coverage received naloxone at the index event (n=0).
- A conservative estimate of 100,000 IOP-RX index events shows naloxone treatment can yield cost savings of ~$140 million for Medicare and ~$83 million for Medicaid. Commercial insurance cost savings cannot be assessed due to the low number of commercial insurance beneficiaries (n=1) that received naloxone in the IOP-RX cohort.

Limitations

- Inherent limitations of claims data analysis include the possibility of incomplete, inaccurate, or missing data. Undetected coding errors could have resulted in the misidentification of RIOP-RX and IOP-RX. Moreover, our use of claims data only allowed for analysis of dispensed prescriptions and diagnoses if a claim was filed. For example, opioid overdose events and opioid use disorder medication dispensing that occurred outside of medical channels are not captured in the data used in this study.
- This study presents illicit-use opioid overdose rates only. Although we limited our cohort population to opioid overdose patients without an opioid prescription, we cannot rule out the possibility that some of our observed opioid overdoses were attributed to the use of prescription opioids that were dispensed to the patient prior to the pre-index period.
- Analysis was restricted to a defined study window with a minimum continuous enrollment of 365 days both pre- and post-index date. Due to this continuous enrollment constraint, our study did not address overdose-induced deaths at the index event.
- Calculated at-index costs were conservative because they do not account for any subsequent overdose events and associated medical encounters for treatment occurring within the same year.
- One needs to use caution and consider the implications of smaller sample size in some of the groups while interpreting these study results.

Conclusions

- Our study provides previously unavailable information on RIOP-RX and IOP-RX rates broken down by patient characteristics, comorbidities, naloxone use, payor type (overall, commercial, Medicare, and Medicaid), and state. States with the highest distribution of RIOP-RX included New Jersey (22.16%), Ohio (13.64%), Tennessee (11.93%), New York (9.66%), and Pennsylvania (8.52%). States with the most IOP-RX cases included Ohio (20.68%), Pennsylvania (11.65%), New Jersey (11.18%), New York (10.17%), and Louisiana (7.16%).
- RIOP-RX and IOP-RX demonstrated a significant economic burden to the U.S. healthcare system, and commercial, Medicare, and Medicaid plan payors. Depending on the payor type, treating 100,000 RIOP-RX annually may cost between ~$375 million (Medicaid) to $657 million (commercial). For IOP-RX these costs range from ~$191 million (Medicaid) to ~$882 million (commercial).
- Overall, the RIOP-RX cohort had higher average index event total costs than the IOP group. These costs differences are likely due to the higher rates of underlying comorbidities among RIOP-RX, resulting in the need for more costly services such as inpatient treatment and advanced medical care.
- Although naloxone use greatly reduced medical costs and healthcare utilization following an illicit-use opioid overdose, this medication remains grossly under-dispensed among RIOP-RX and IOP-RX.
- Given increasingly constrained healthcare budgets, higher rates of naloxone co-prescribing may help reduce the economic burden associated with treating RIOP-RX and IOP-RX populations.

Future Research

- Further studies are needed to explore the impact of naloxone use on clinical and other benefits in RIOP-RX and IOP-RX.
- Future research should explore the relationship between naloxone use, lower medical costs, and decreased utilization of healthcare resources among RIOP-RX and IOP-RX.
- Additional studies are needed to assess the benefits of naloxone prescribing among RIOP-RX and IOP-RX using data from states that have implemented co-prescribing policies.
- Future studies should examine why naloxone is currently prescribed at such a low rate among illicit opioid users with respiratory comorbidity. Once these naloxone prescribing barriers are identified, we must explore initiatives to improve naloxone prescriptions among this vulnerable population.

Acknowledgements

The authors of this presentation would like to thank ScioScientific and the Power BI team from Emergent BioSolutions for their contributions to this poster.

References

1. Substance Abuse and Mental Health Services Administration. (2021). *Key substance use and mental health indicators in the United States: Results from the 2020 National Survey on Drug Use and Health* (HHS Publication No. PEP21-07-01-003, NSDUH Series H-56). Rockville, MD: Center for Behavioral Health Statistics and Quality, Substance Abuse and Mental Health Services Administration.
2. National Institute on Drug Abuse. Trends and Statistics: Overdose Death Rates. https://nida.nih.gov/drug-topics/trends-statistics/overdose-death-rates; retrieved January 27, 2022.
3. Premier Inc. "Opioid Overdoses Costing U.S. Hospitals an Estimated $11 Billion Annually," news release, Jan. 3, 2019. https://www.premierinc.com/newsroom/press-releases/opioid-overdoses-costing-u-s-hospitals-an-estimated-11-billion-annually; retrieved February 8, 2022.
4. Hulin J, Brodie A, Stevens J, Mitchell C. Prevalence of respiratory conditions among people who use illicit opioids: a systematic review. *Addiction*. 2020 May;115(5):832-849.
5. Tas B, Kalk N, Cho P, et al. Respiratory depression in opioid dependent chronic obstructive pulmonary disease patients. *Thorax* 2021;76:A113-A114.
6. Crystal S, Nowels M, Olfson M, Samples H, Williams AR, Treitler P. Medically treated opioid overdoses among New Jersey Medicaid beneficiaries: Rapid growth and complex comorbidity amid growing fentanyl penetration. *J Subst Abuse Treat*. 2021 Dec;131:108546.
7. Au VYO, Rosic T, Sanger N, Hillmer A, Chawar C, Worster A, Marsh DC, Thabane L, Samaan Z. Factors associated with opioid overdose during medication-assisted treatment: How can we identify individuals at risk? *Harm Reduct J*. 2021 Jul 8;18(1):71.
8. Tas B, Kalk NJ, Lozano-García M, Rafferty GF, Cho P, Kelleher M, Moxham J, Strang J, Jolley CJ. Undetected Respiratory Depression in People with Opioid Use Disorder. *Drug Alcohol Depend*. 2022 Mar 10;234:109401.
9. Centers for Disease Control and Prevention. Life-Saving Naloxone from Pharmacies. https://www.cdc.gov/vitalsigns/naloxone/index.html; retrieved March 9, 2022.
10. McDonald R, Campbell ND, Strang J. Twenty years of take-home naloxone for the prevention of overdose deaths from heroin and other opioids-Conception and maturation. *Drug Alcohol Depend*. 2017 Sep 1;178:176-187.
11. Bennett AS, Scheidell J, Bowles JM, Khan M, Roth A, Hoff L, Marini C, Elliott L. Naloxone protection, social support, network characteristics, and overdose experiences among a cohort of people who use illicit opioids in New York City. *Harm Reduct J*. 2022 Mar 4;19(1):20.
12. Optum's de-identified Clinformatics® Data Mart Database (2007-2021).