Real-World Opioid Overdose Trends, Costs, and Comorbidities in Commercial, Managed Medicare, and Managed Medicaid Patients in the United States (2016-2019)

Arun Changolkar, MBA, PhD, MSc[1]; Mohan K. Sindhwani, MD[2]; Shaheen Kaplan, PharmD, MBA[2]; Gay Owens, PharmD, MBA[2]

[1]Emergent BioSolutions, Philadelphia, PA, USA, [2]Emergent BioSolutions, Gaithersburg, MD USA

Background

Opioid Overdose in the United States

- In 2020, nearly 10 million Americans misused opioids and 2.7 million had an opioid use disorder.[1]
- Three-quarters of all fatal drug overdoses in 2020 involved an illicit or prescription opioid drug.[2]
 - 68,630 overdose deaths involved any opioid drug.
 - 56,516 deaths involved synthetic opioids (primarily fentanyl).
 - 16,416 deaths involved prescription opioids.
 - 13,165 deaths involved heroin.
- Opioid overdoses were responsible for 1 in every 313 emergency department visits in 2020.[3]
- Opioid overdose patients account for more than $11 billion in annual hospital costs nationwide.[4]
- Opioid use disorder and fatal opioid overdoses cost the United States more than $1 trillion a year.[5]

Naloxone

- Naloxone is a prescription medication that counters the effects of opioid overdose, helping to restore normal breathing and prevent death in an overdose victim.
- According to current opioid prescribing guidelines from the Centers for Disease Control and Prevention (CDC), naloxone should be co-prescribed to all individuals at risk for opioid overdose.[6]

Objectives

- To assess demographic and clinical characteristics of opioid overdose patients (OOP) in the United States from 2016 to 2019 by payor (commercial, Medicare, Medicaid).
- To examine baseline comorbidities of OOP by payor and year.
- To analyze healthcare expenditure of OOP by payor and year.

Methods

Study Design

- Retrospective cohort study.

Data Source

- Optum's Clinformatics® Data Mart (CDM) that is derived from administrative health claims for members of large Commercial, Medicare Advantage, and Managed Medicaid health plans.[7]

Patient Inclusion Criteria

- Medical claim for an opioid overdose diagnosis (prescription or illicit) between January 1, 2016 through December 31, 2019.
- ≥18 years of age with continuous medical and pharmacy coverage for a minimum of 30 days at index event (first claim date for opioid overdose).

Analysis

- International Classification of Disease-10-Clinical Modification (ICD-10-CM) codes (corresponding to poisoning by opium/ opioids, heroin, methadone, and synthetic or unspecified narcotics) were used to identify OOP.
- OOP were analyzed by patient characteristics, comorbidities, state, medical costs, payor type (commercial, Medicare, and Medicaid), and year.
- Total and disease-specific per member per month costs (PMPMC) were analyzed annually by payor (commercial, Medicare, and Medicaid).
 - Disease-specific medical costs included claims related to opioid poisoning.
 - Disease-specific pharmacy costs included claims for naloxone (alone or combination drug).
- OOP comorbidities were characterized by payor (commercial, Medicare, and Medicaid) and year using the Charlson Comorbidity Index (CCI) and Agency for Healthcare Research and Quality (AHRQ1-3) comorbidity indexes.

Results

Figure 1. Annual Trends in Age Group Distribution of Opioid Overdose Patients, by Payor (2016-2019).

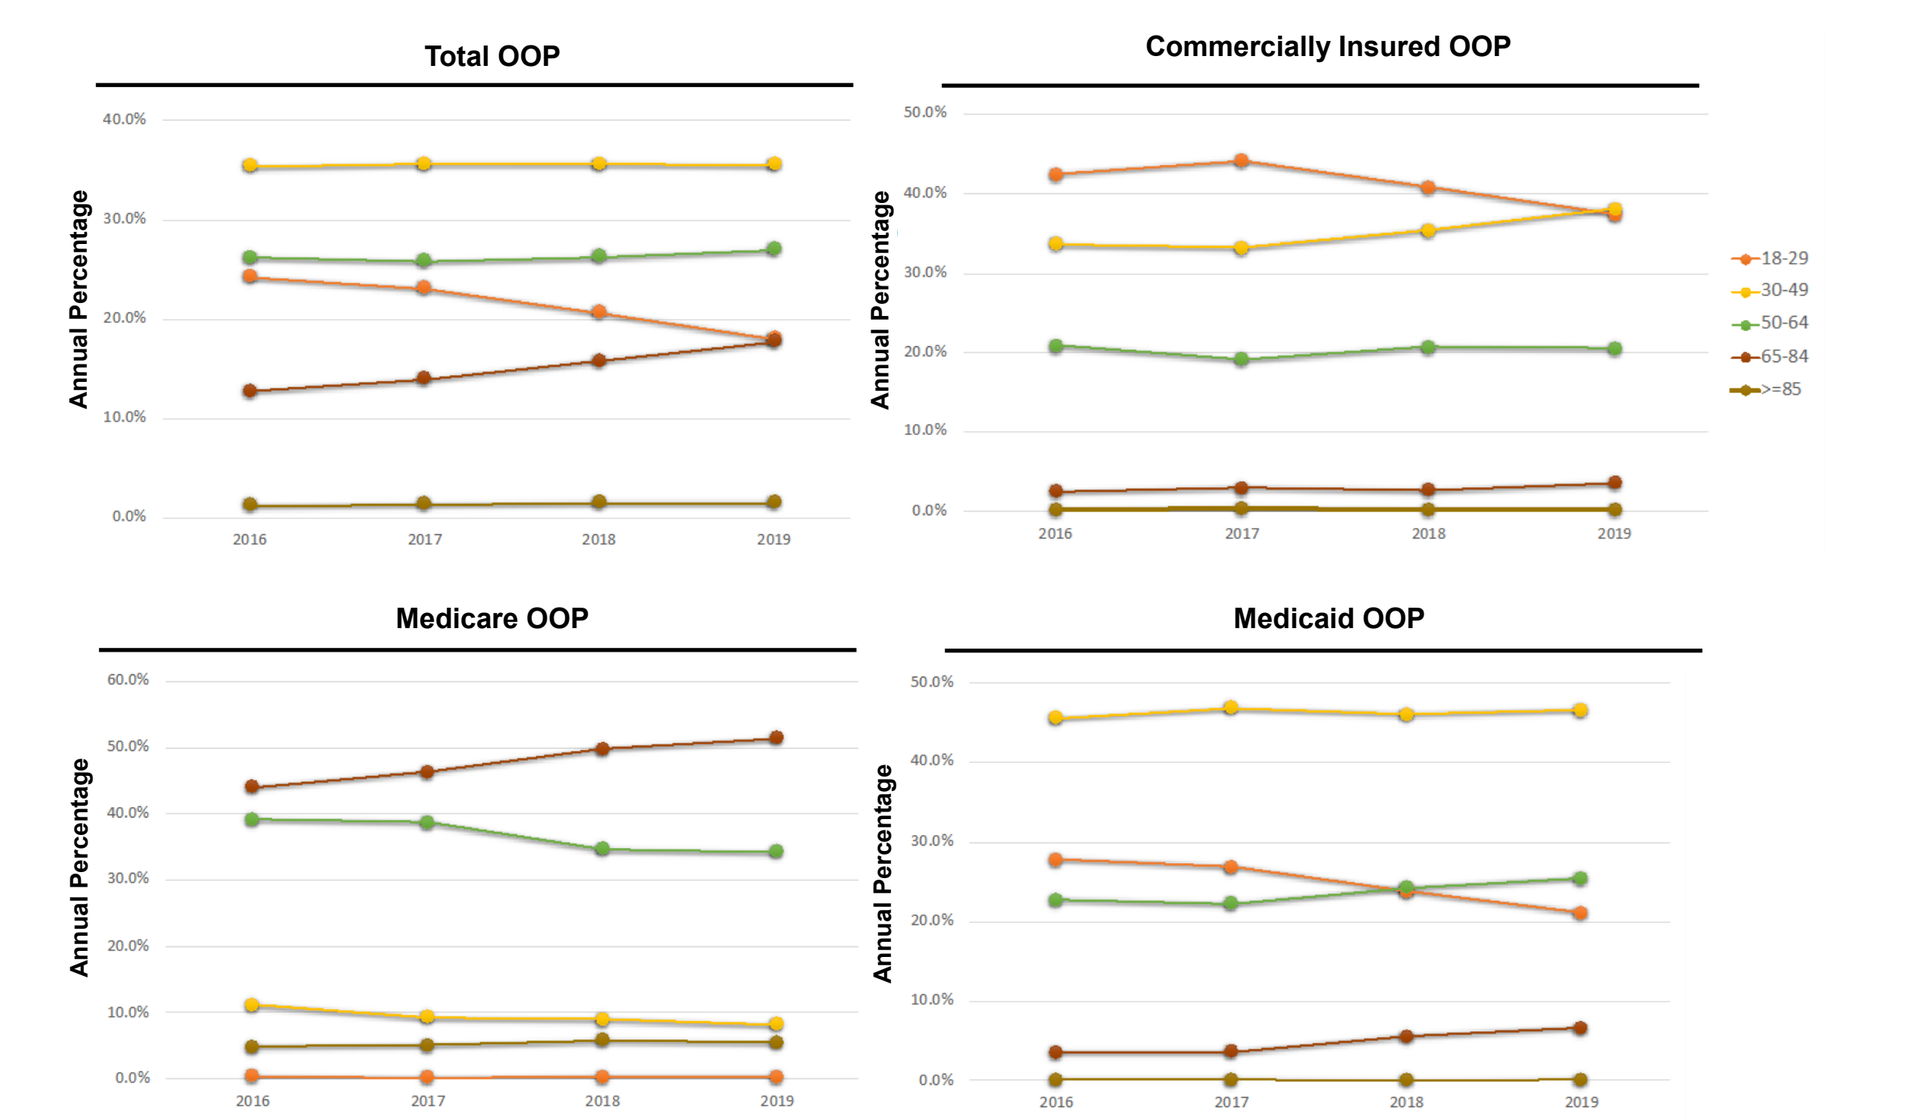
Total OOP
Commercially Insured OOP
Medicare OOP
Medicaid OOP
Annual Percentage
18-29
30-49
50-64
65-84
>=85

Figure 2. Increasing Trends in Opioid Overdose Patients Aged 50 Years or Greater, by Payor (2016-2019).

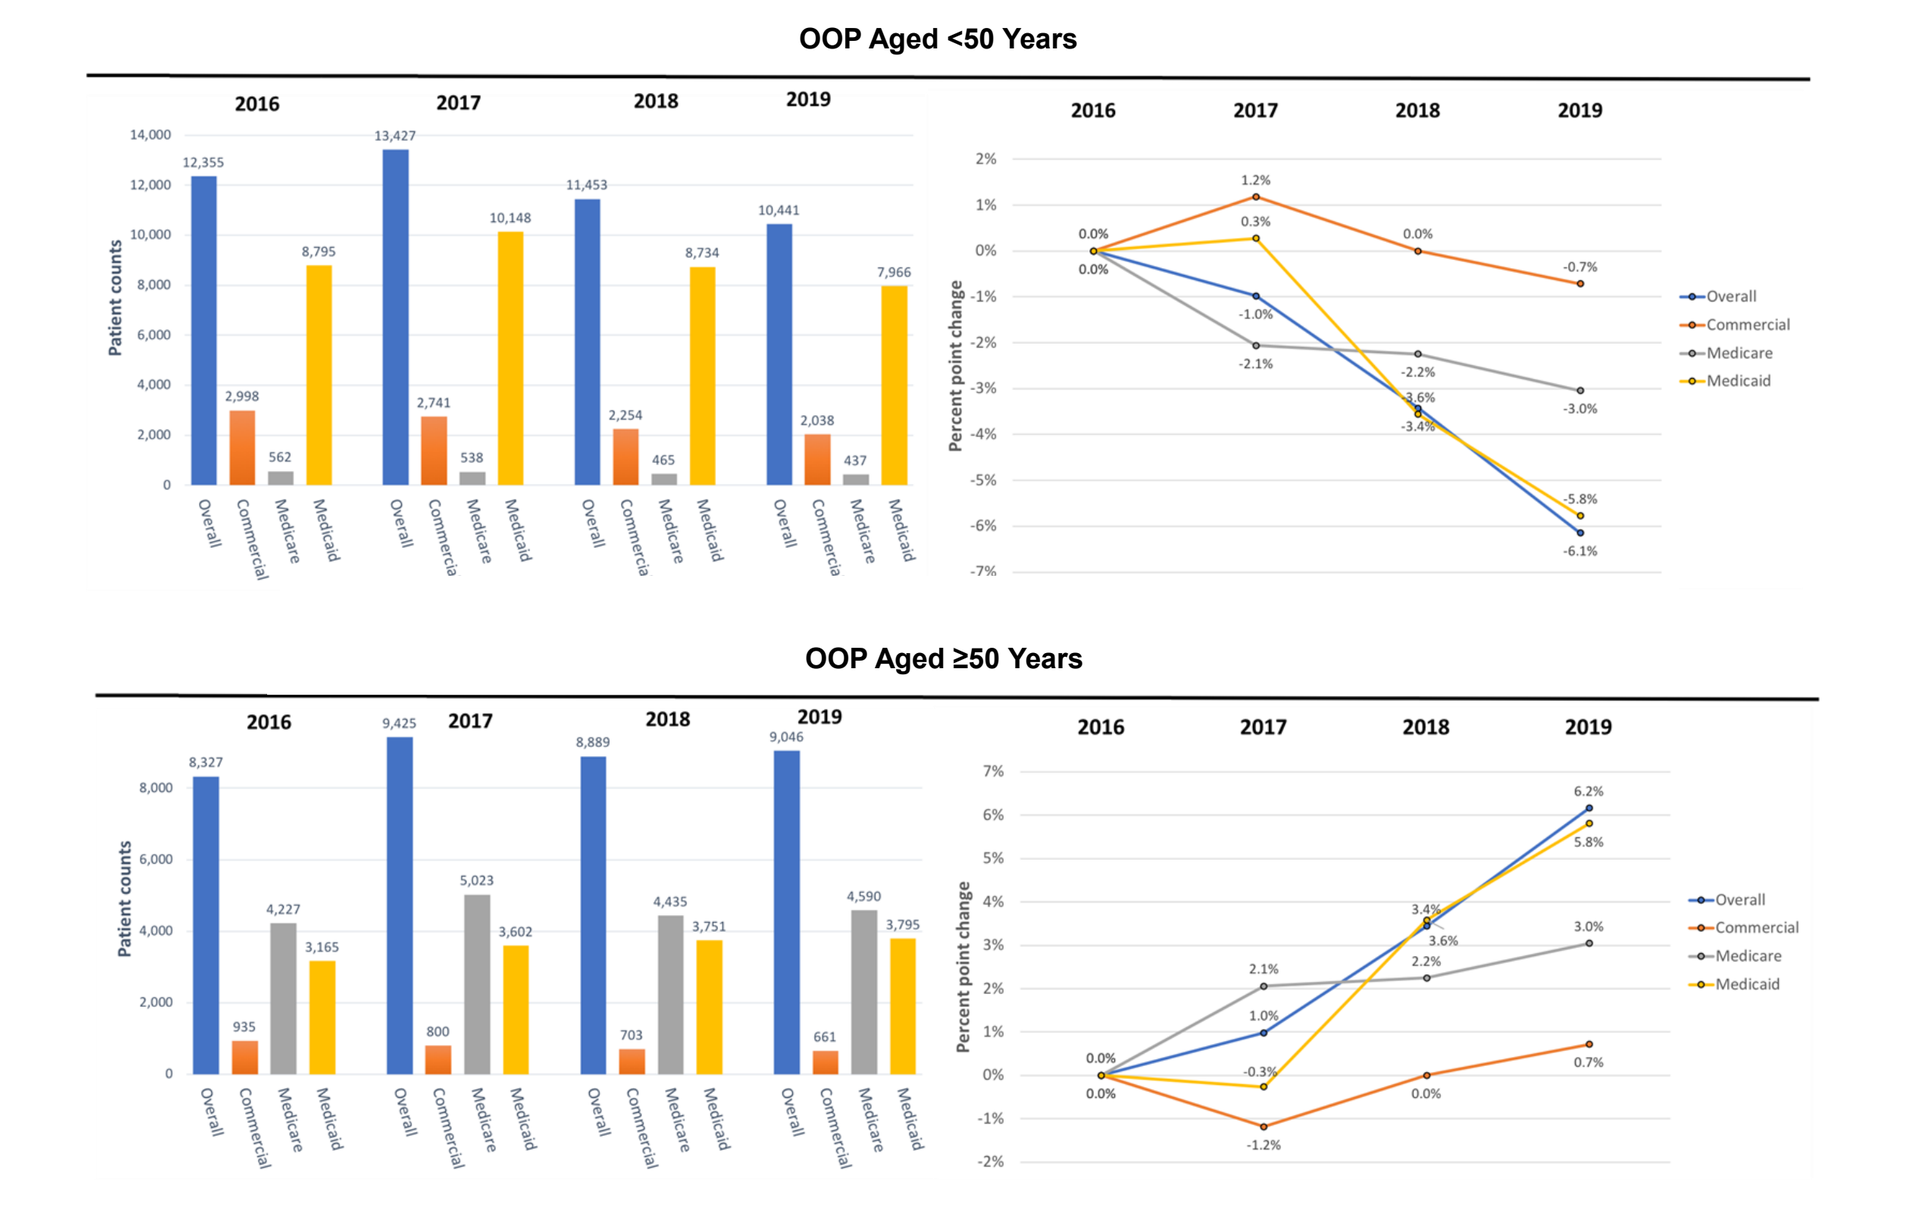
OOP Aged <50 Years
OOP Aged ≥50 Years
Patient counts
Percent point change
Overall
Commercial
Medicare
Medicaid

Figure 3. Annual Trends in Mean Age of Opioid Overdose Patients, by Payor (2016-2019).

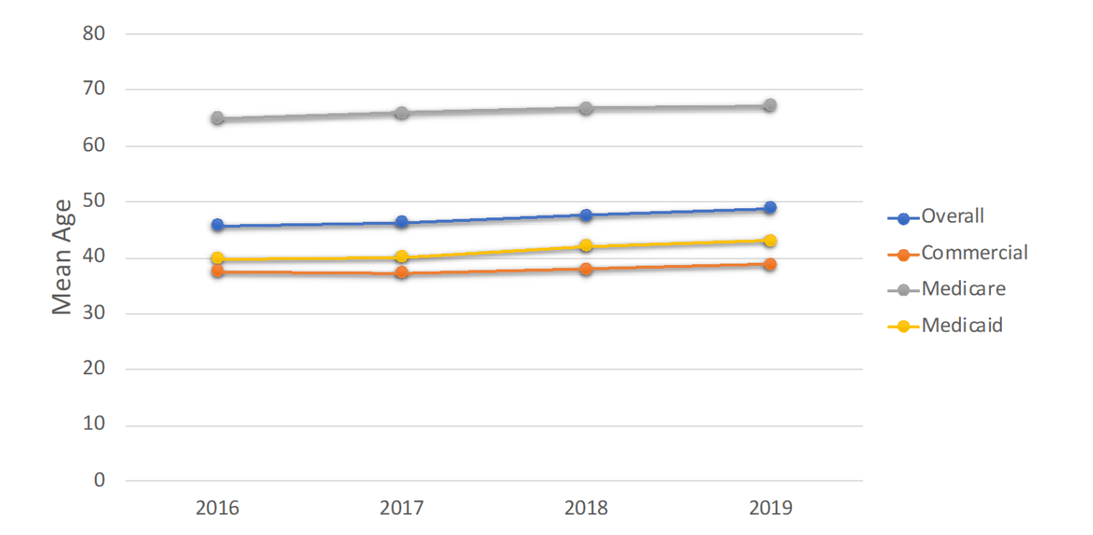
Mean Age
Overall
Commercial
Medicare
Medicaid

Figure 4. Annual Trends in Gender Distribution of Opioid Overdose Patients, by Payor (2016-2019).

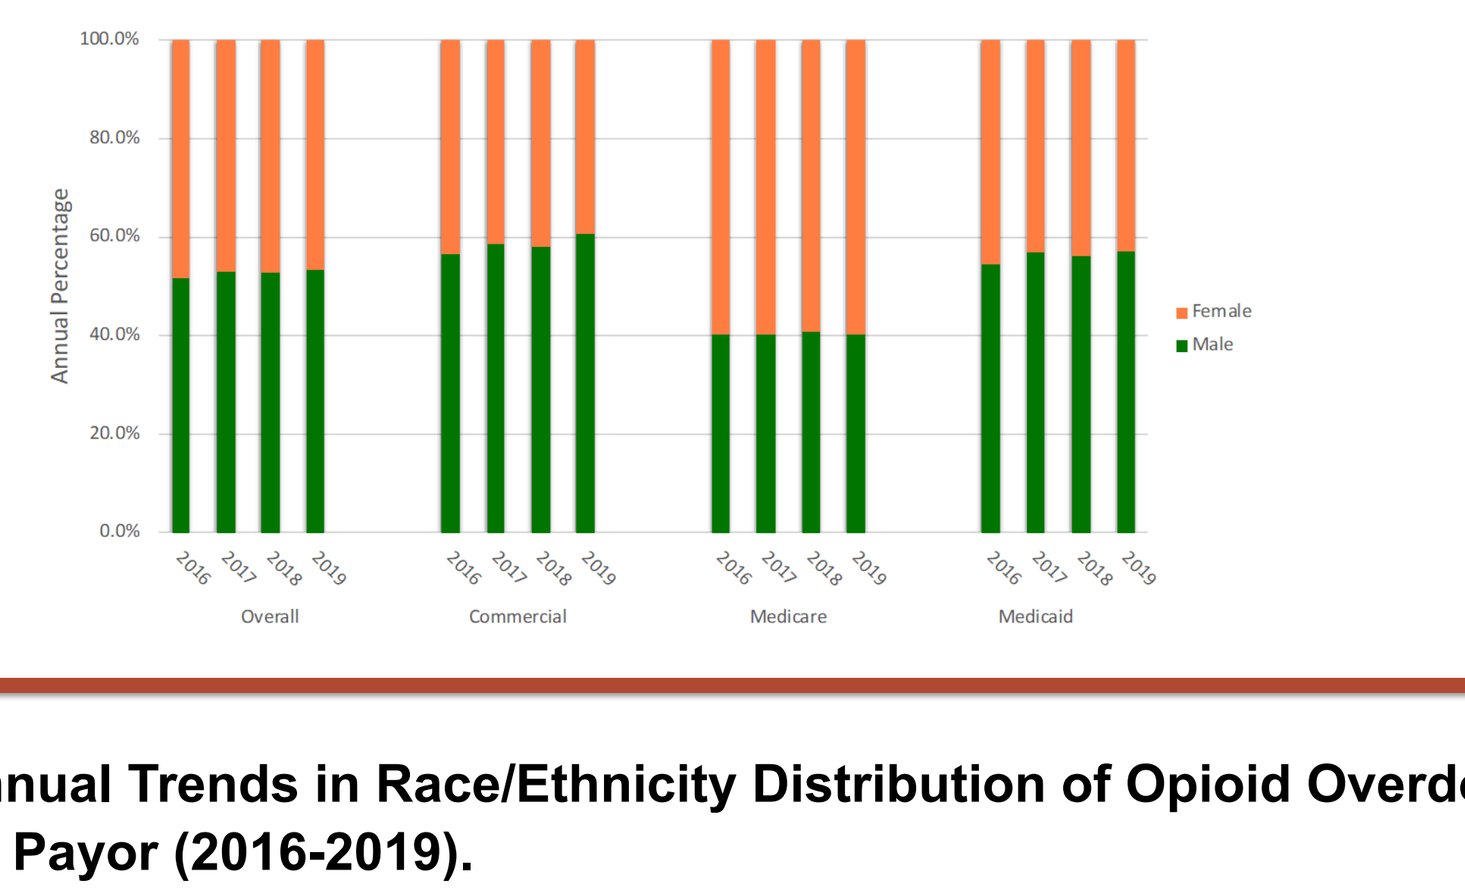
Annual Percentage
Female
Male
Overall
Commercial
Medicare
Medicaid

Figure 5. Annual Trends in Race/Ethnicity Distribution of Opioid Overdose Patients, by Payor (2016-2019).

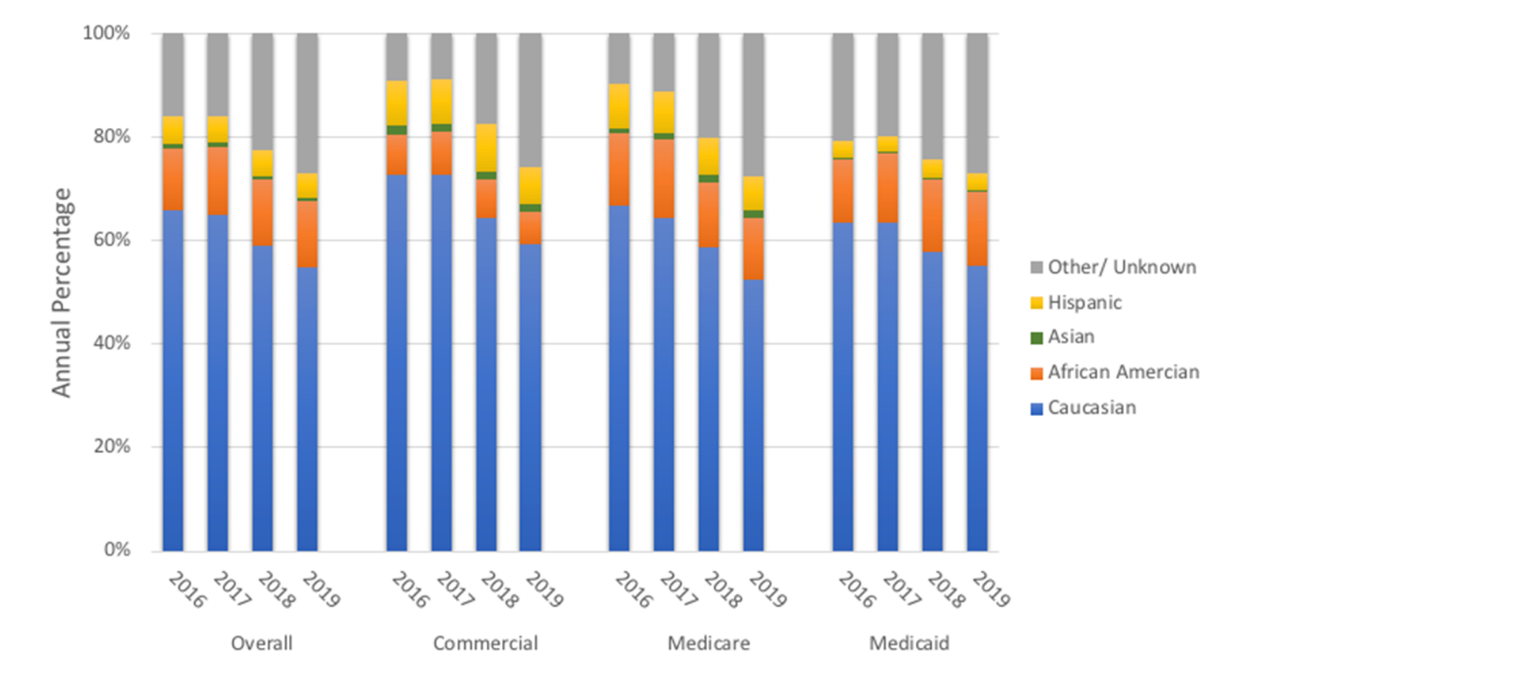
Annual Percentage
Other/ Unknown
Hispanic
Asian
African American
Caucasian
Overall
Commercial
Medicare
Medicaid

Figure 6. Annual Trends in State-Level Distribution of Opioid Overdose Patients, by Payor (2016-2019).

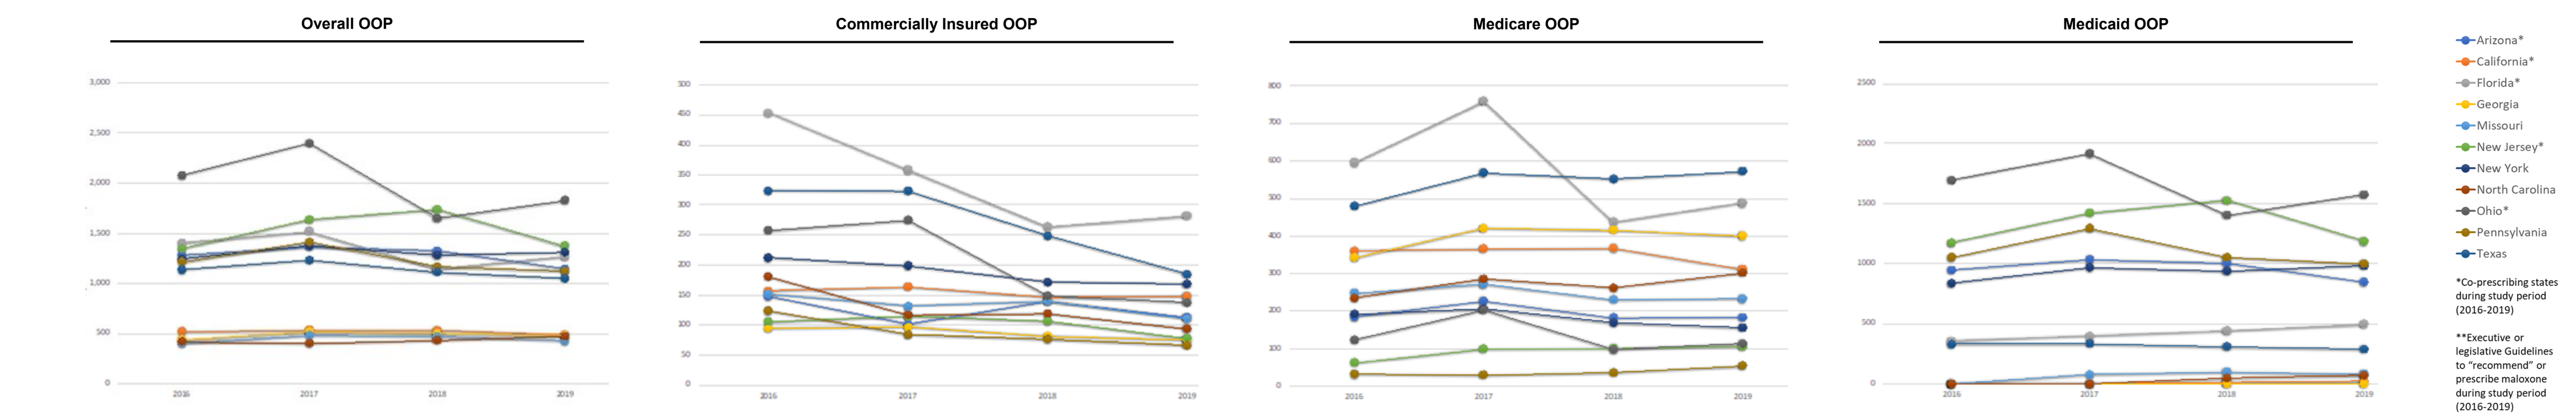
Overall OOP
Commercially Insured OOP
Medicare OOP
Medicaid OOP
Arizona*
California*
Florida*
Georgia
Missouri
New Jersey*
New York
North Carolina
Ohio*
Pennsylvania
Texas
*Co-prescribing states during study period (2016-2019)
**Executive or legislative Guidelines to "recommend" or prescribe maloxone during study period (2016-2019)

Figure 7. Annual Trends in Average CCI Score of Opioid Overdose Patients, by Payor (2016-2019).

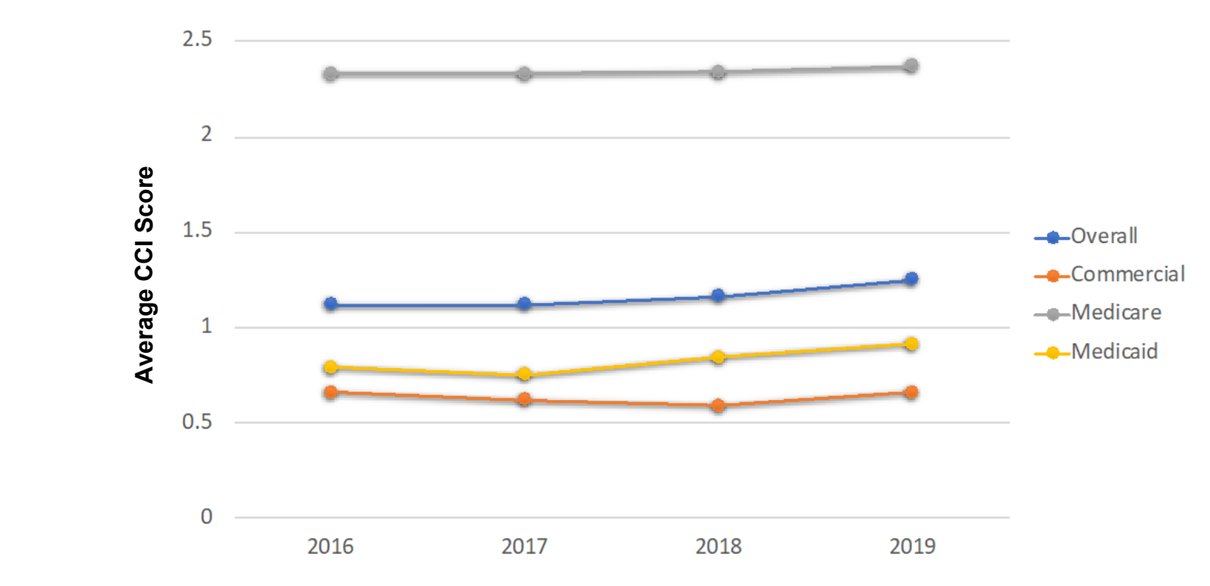
Average CCI Score
Overall
Commercial
Medicare
Medicaid

Figure 8. Annual Trends in Opioid Overdose Patients with ≥1 CCI Comorbidity, by Payor (2016-2019).

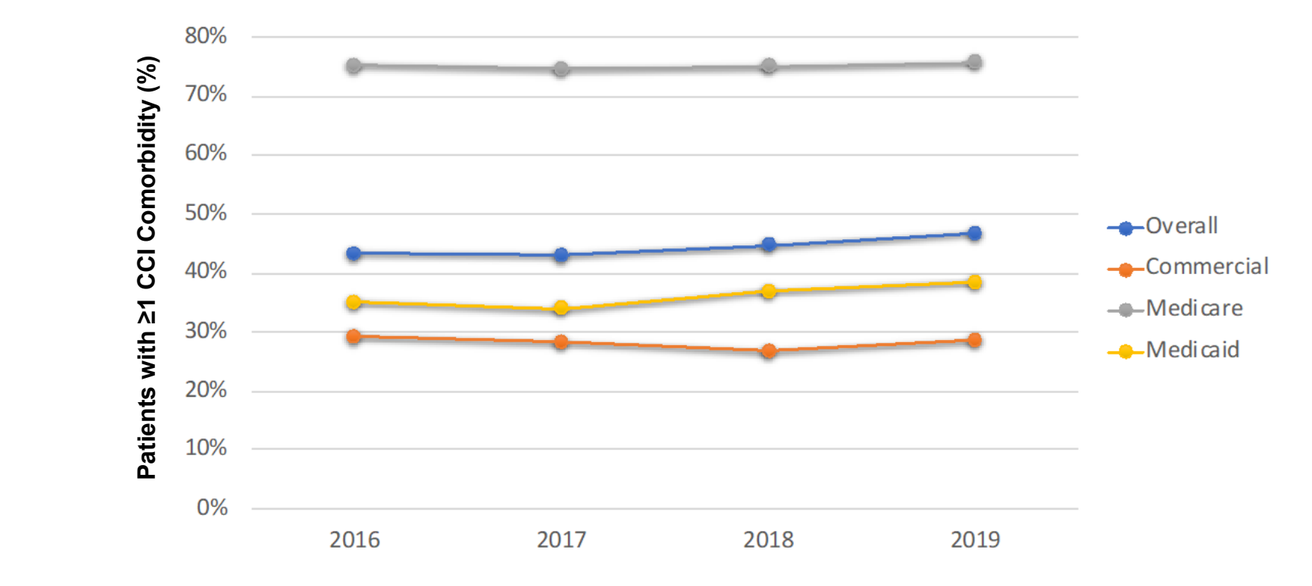
Patients with ≥1 CCI Comorbidity (%)
Overall
Commercial
Medicare
Medicaid

Figure 9. Annual Trends in CCI Comorbidities among Opioid Overdose Patients, by Payor (2016-2019).

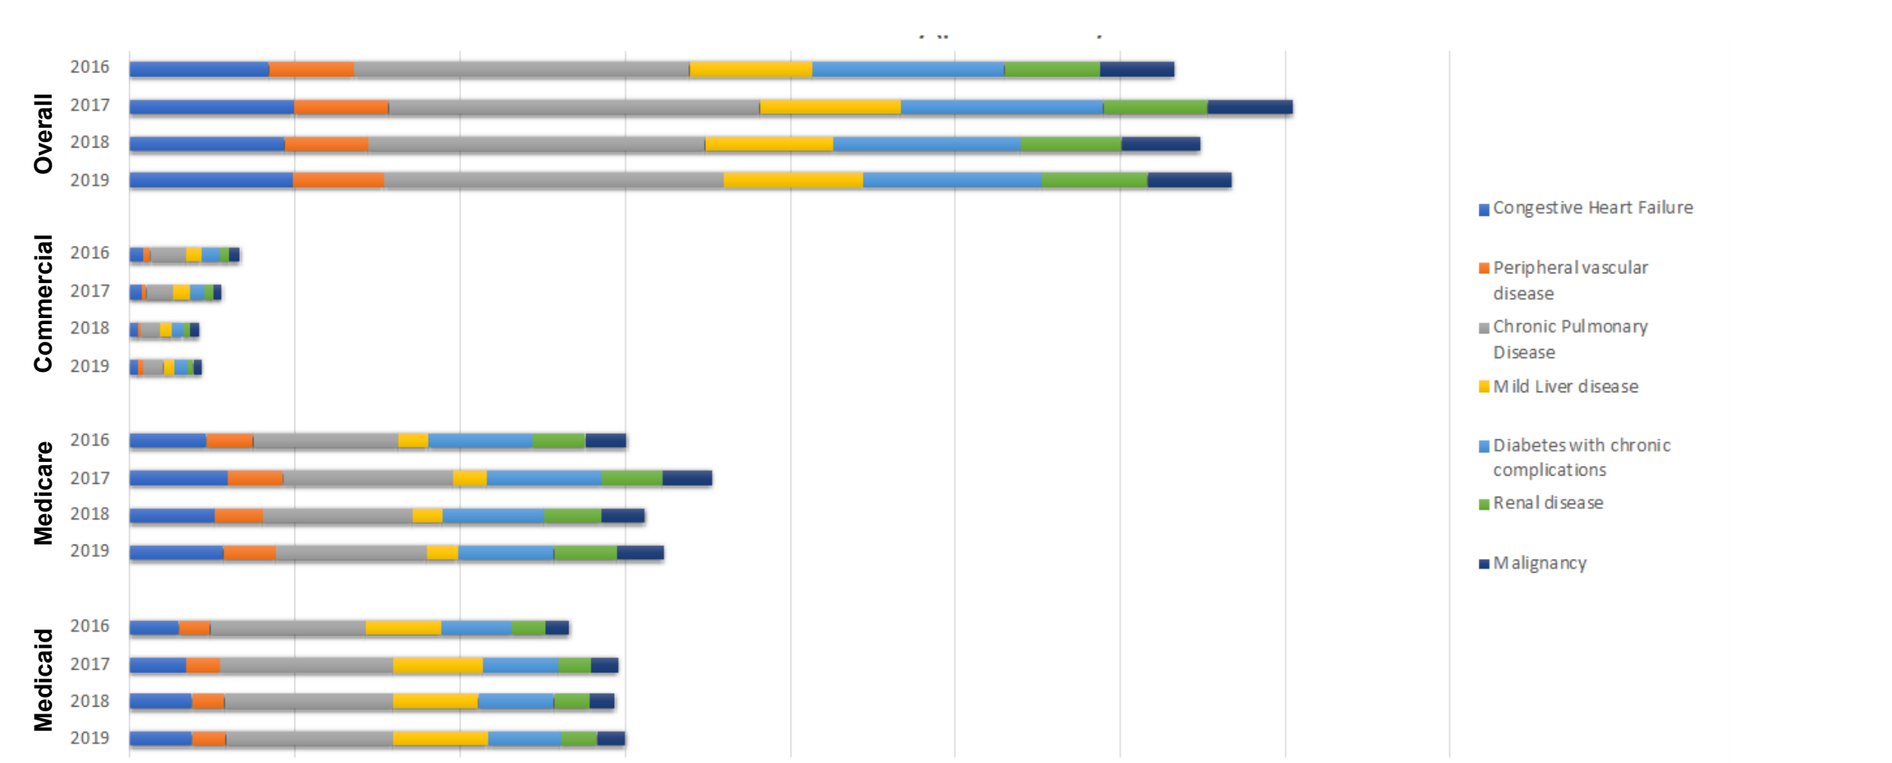
Overall
Commercial
Medicare
Medicaid
Congestive Heart Failure
Peripheral vascular disease
Chronic Pulmonary Disease
Mild Liver disease
Diabetes with chronic complications
Renal disease
Malignancy

	Overall				Commercial				Medicare				Medicaid			
	2016	2017	2018	2019	2016	2017	2018	2019	2016	2017	2018	2019	2016	2017	2018	2019
Congestive heart failure	1,688	1,999	1,869	1,972	158	140	91	97	930	1,178	1,030	1,125	600	681	748	750
Peripheral vascular disease	1,041	1,142	1,031	1,113	91	58	41	63	571	675	586	642	379	409	404	408
Chronic pulmonary disease	4,058	4,493	4,073	4,122	428	333	232	242	1,750	2,055	1,807	1,843	1,880	2,105	2,034	2,037
Mild liver disease	1,481	1,716	1,553	1,685	192	200	136	144	372	427	376	382	917	1,089	1,041	1,159
Diabetes with chronic complications	2,338	2,448	2,281	2,172	226	167	150	147	1,264	1,376	1,221	1,148	848	905	910	877
Renal disease	1,156	1,265	1,214	1,271	104	107	87	74	645	753	686	765	407	405	441	432
Malignancy	913	1,043	953	1,022	131	116	96	101	488	593	540	578	294	334	317	343

Figure 10. Annual Trends in AHRQ1 Comorbidities among Opioid Overdose Patients, by Payor (2016-2019).

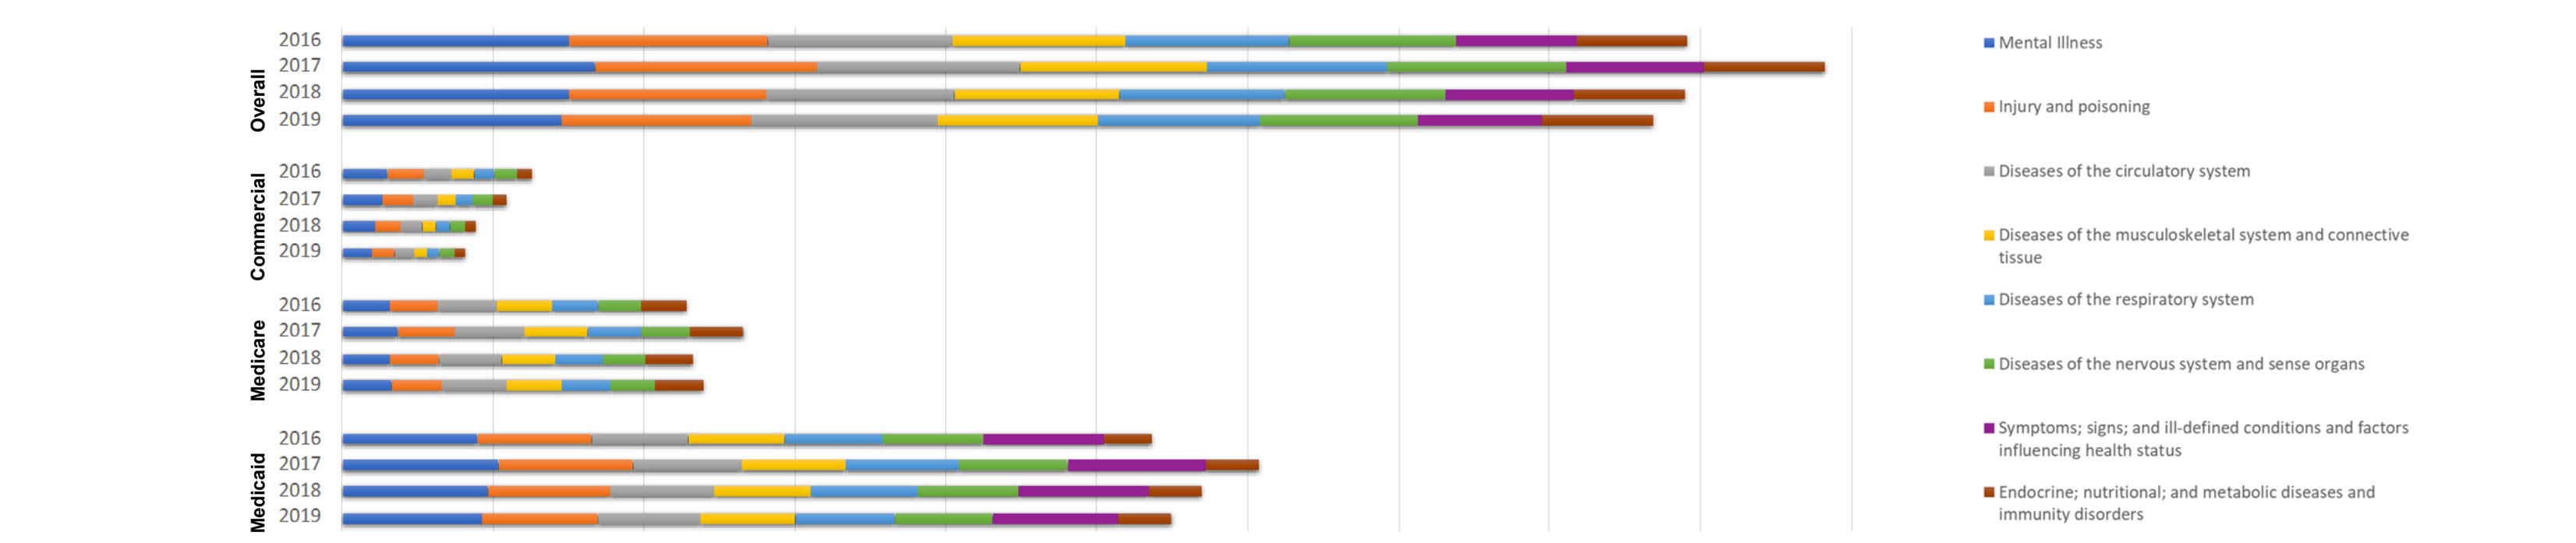
Overall
Commercial
Medicare
Medicaid
Mental Illness
Injury and poisoning
Diseases of the circulatory system
Diseases of the musculoskeletal system and connective tissue
Diseases of the respiratory system
Diseases of the nervous system and sense organs
Symptoms; signs; and ill-defined conditions and factors influencing health status
Endocrine; nutritional; and metabolic diseases and immunity disorders

	Overall				Commercial				Medicare				Medicaid			
	2016	2017	2018	2019	2016	2017	2018	2019	2016	2017	2018	2019	2016	2017	2018	2019
Mental Illness	15,091	16,720	15,007	14,556	3,010	2,642	2,192	1,978	3,142	3,673	3,145	3,300	8,939	10,405	9,670	9,278
Injury and poisoning	13,153	14,786	13,118	12,525	2,359	2,066	1,708	1,511	3,188	3,783	3,280	3,321	7,606	8,937	8,130	7,693
Diseases of the circulatory system	12,271	13,435	12,430	12,355	1,917	1,650	1,401	1,288	3,988	4,679	4,181	4,294	6,366	7,106	6,848	6,773
Diseases of the musculoskeletal system and connective tissue	11,423	12,333	10,929	10,699	1,439	1,191	924	809	3,571	4,166	3,553	3,635	6,413	6,976	6,452	6,255
Diseases of the respiratory system	10,894	12,050	11,029	10,739	1,321	1,117	890	877	3,038	3,510	3,122	3,201	6,535	7,423	7,017	6,661
Diseases of the nervous system and sense organs	11,020	11,841	10,585	10,379	1,511	1,310	1,034	938	2,845	3,256	2,799	2,937	6,664	7,275	6,752	6,504
Symptoms; signs; and ill-defined conditions and factors influencing health status	8,021	9,104	8,582	8,270									8,021	9,104	8,582	8,270
Endocrine; nutritional; and metabolic diseases and immunity disorders	7,222	8,031	7,367	7,434	1,023	893	702	703	3,038	3,570	3,133	3,229	3,161	3,568	3,532	3,502

Figure 11. Annual Trends in AHRQ2 Comorbidities among Opioid Overdose Patients, by Payor (2016-2019).

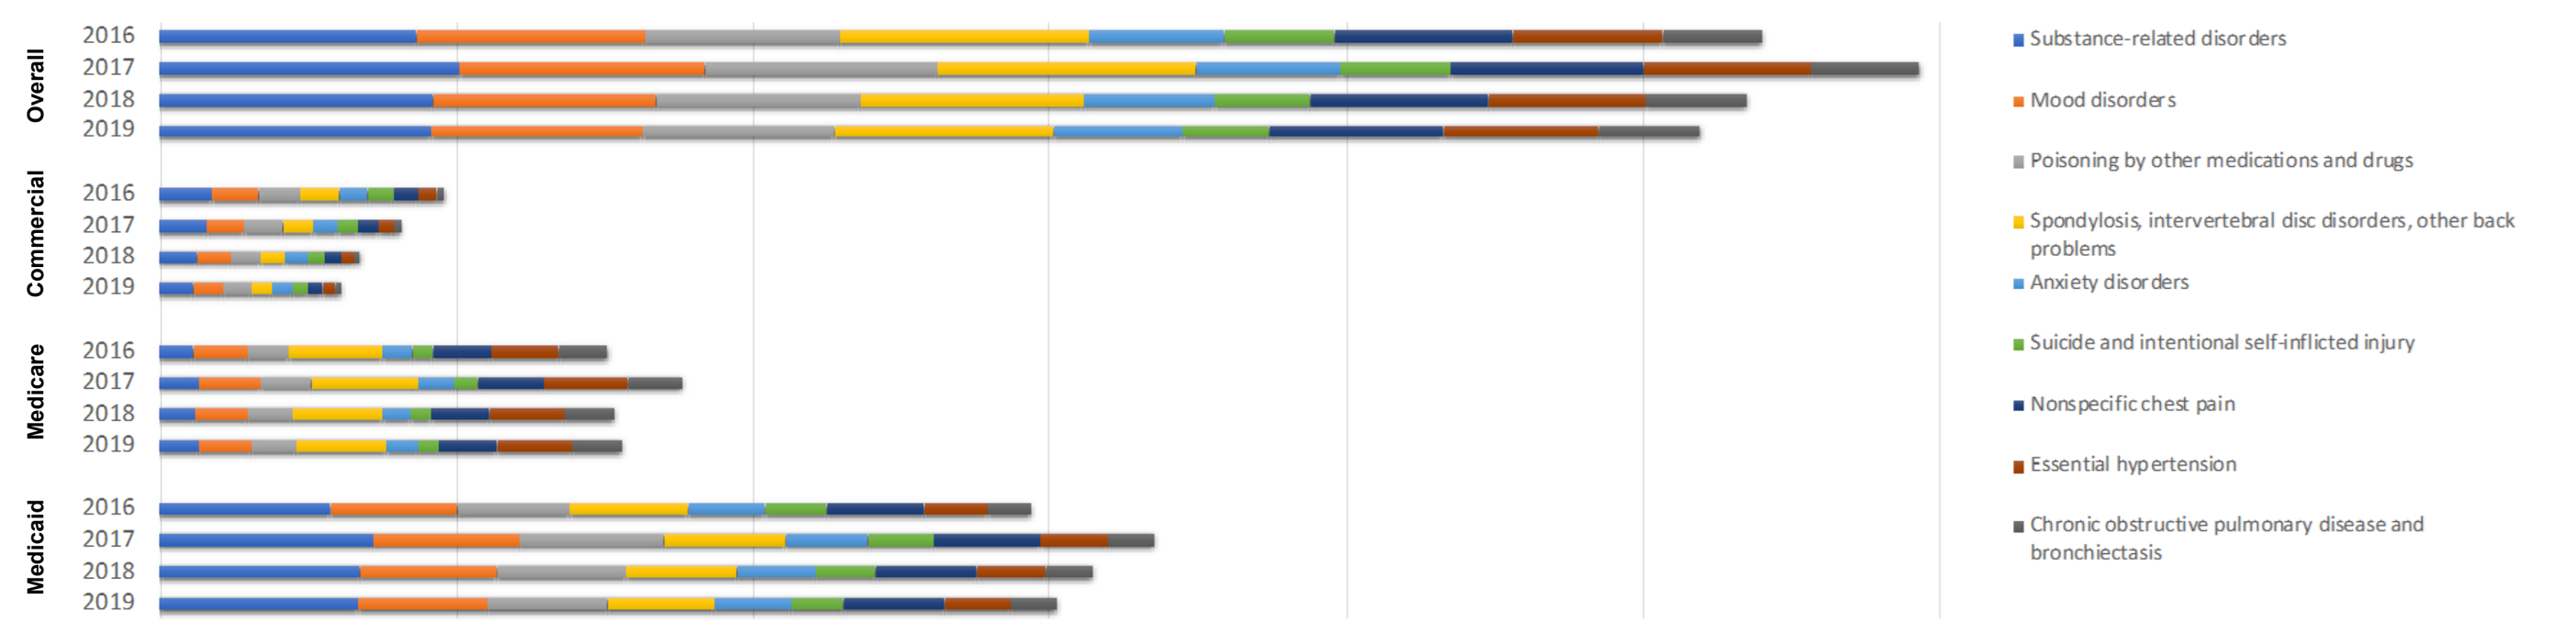
Overall
Commercial
Medicare
Medicaid
Substance-related disorders
Mood disorders
Poisoning by other medications and drugs
Spondylosis, intervertebral disc disorders, other back problems
Anxiety disorders
Suicide and intentional self-inflicted injury
Nonspecific chest pain
Essential hypertension
Chronic obstructive pulmonary disease and bronchiectasis

	Overall				Commercial				Medicare				Medicaid			
	2016	2017	2018	2019	2016	2017	2018	2019	2016	2017	2018	2019	2016	2017	2018	2019
Substance-related disorders	8,632	10,121	9,207	9,118	1,761	1,569	1,244	1,131	1,118	1,340	1,193	1,283	5,753	7,212	6,770	6,704
Mood disorders	7,703	8,282	7,509	7,164	1,562	1,274	1,124	1,021	1,848	2,073	1,767	1,807	4,293	4,935	4,618	4,336
Poisoning by other medications and drugs	6,616	7,823	6,908	6,463	1,430	1,282	1,058	940	1,404	1,704	1,486	1,481	3,782	4,837	4,364	4,042
Spondylosis, intervertebral disc disorders, other back problems	8,382	8,712	7,541	7,370	1,296	1,046	812	705	3,118	3,585	3,042	3,068	3,968	4,081	3,687	3,597
Anxiety disorders	4,568	4,870	4,378	4,358	954	819	728	677	997	1,226	962	1,054	2,617	2,825	2,688	2,627
Suicide and intentional self-inflicted injury	3,692	3,700	3,253	2,935	876	708	602	526	731	795	674	680	2,085	2,197	1,977	1,729
Nonspecific chest pain	6,021	6,519	5,984	5,877	808	687	572	502	1,930	2,241	2,001	1,971	3,283	3,591	3,411	3,404
Essential hypertension	5,057	5,645	5,327	5,251	627	527	445	426	2,316	2,808	2,532	2,564	2,114	2,310	2,350	2,261
Chronic obstructive pulmonary disease and bronchiectasis	3,363	3,658	3,388	3,432	289	228	135	157	1,622	1,873	1,675	1,705	1,452	1,557	1,578	1,570

Figure 12. Annual Trends in AHRQ3 Comorbidities among Opioid Overdose Patients, by Payor (2016-2019).

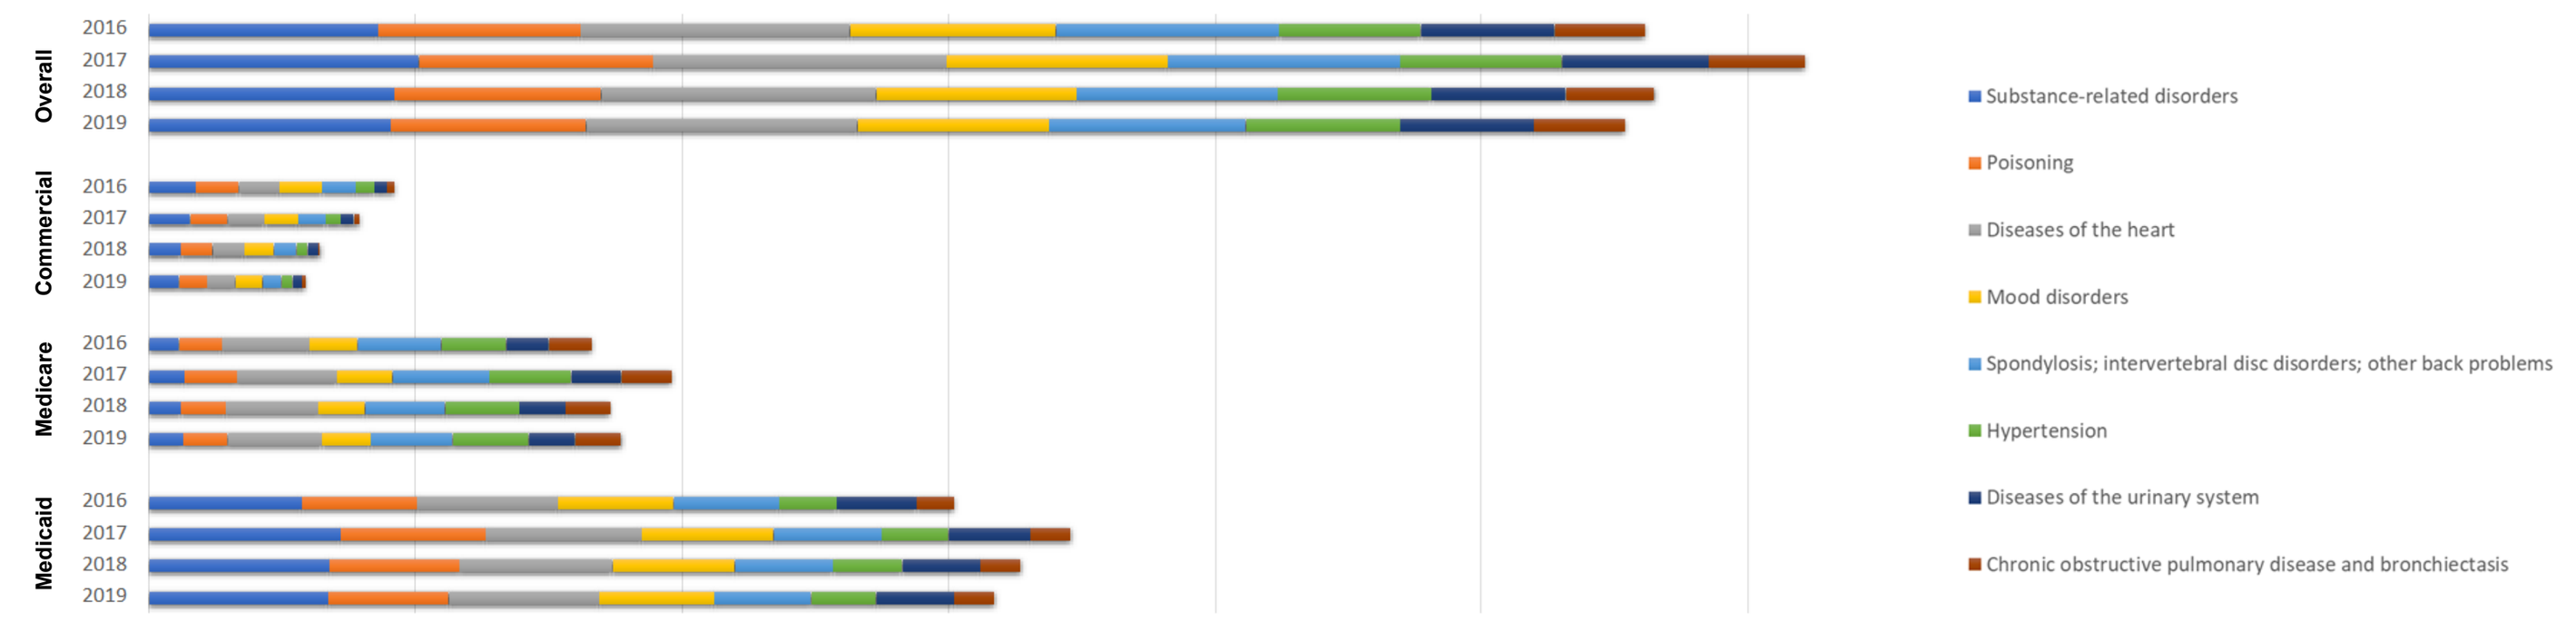
Overall
Commercial
Medicare
Medicaid
Substance-related disorders
Poisoning
Diseases of the heart
Mood disorders
Spondylosis; intervertebral disc disorders; other back problems
Hypertension
Diseases of the urinary system
Chronic obstructive pulmonary disease and bronchiectasis

	Overall				Commercial				Medicare				Medicaid			
	2016	2017	2018	2019	2016	2017	2018	2019	2016	2017	2018	2019	2016	2017	2018	2019
Substance-related disorders	8,632	10,121	9,207	9,118	1,761	1,569	1,244	1,131	1,118	1,340	1,193	1,283	5,753	7,212	6,770	6,704
Poisoning	7,576	8,816	7,771	7,275	1,603	1,417	1,164	1,038	1,643	1,982	1,702	1,699	4,330	5,417	4,905	4,538
Diseases of the heart	10,109	11,026	10,335	10,225	1,570	1,380	1,167	1,075	3,253	3,779	3,446	3,512	5,286	5,867	5,722	5,638
Mood disorders	7,703	8,282	7,509	7,164	1,562	1,274	1,124	1,021	1,848	2,073	1,767	1,807	4,293	4,935	4,618	4,336
Spondylosis; intervertebral disc disorders; other back problems	8,382	8,712	7,541	7,370	1,296	1,046	812	705	3,118	3,585	3,042	3,068	3,968	4,081	3,687	3,597
Hypertension	5,333	6,082	5,752	5,794	648	557	465	458	2,460	3,065	2,755	2,871	2,225	2,460	2,532	2,465
Diseases of the urinary system	5,028	5,442	5,013	5,005	506	442	366	324	1,585	1,898	1,707	1,763	2,937	3,102	2,940	2,918
Chronic obstructive pulmonary disease and bronchiectasis	3,363	3,658	3,388	3,432	289	228	135	157	1,622	1,873	1,675	1,705	1,452	1,557	1,578	1,570

Figure 13. Annual Trends in Total Healthcare Expenditure (Per Member Per Month Costs) among Opioid Overdose Patients, by Payor (2016-2019).

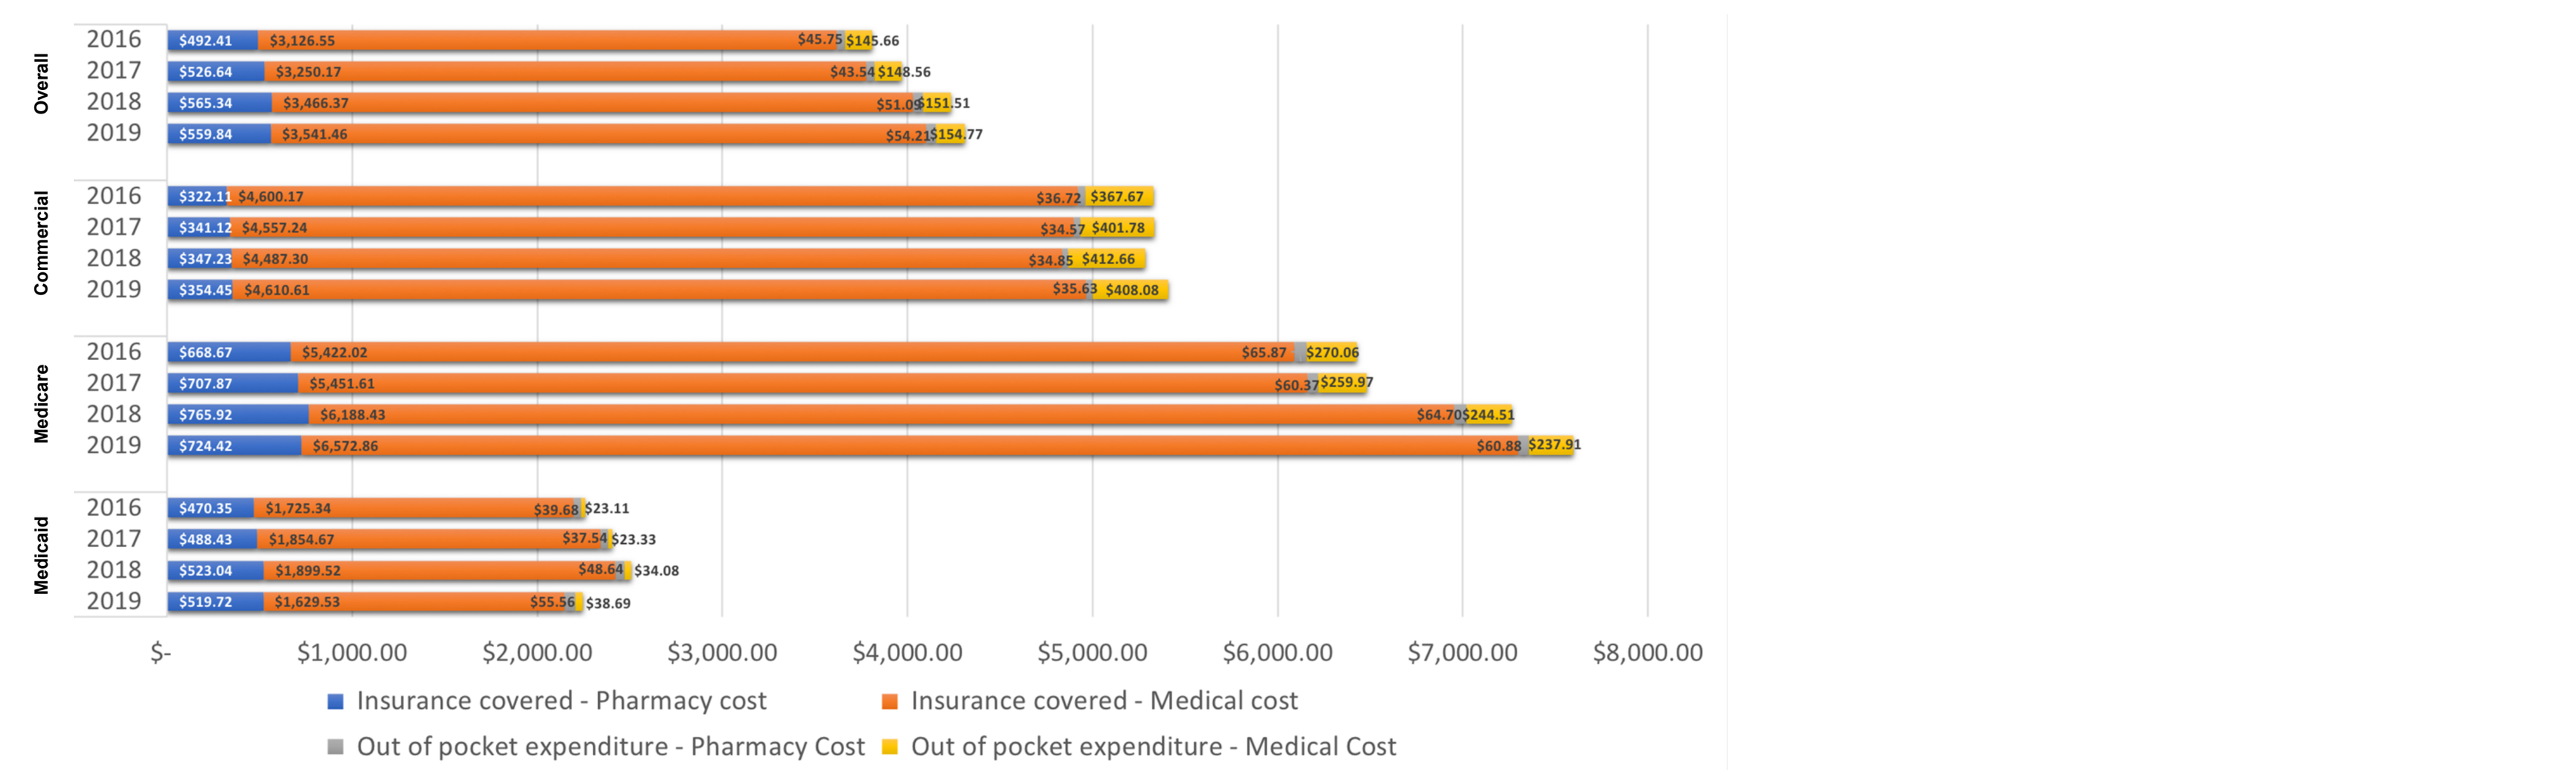
Overall
Commercial
Medicare
Medicaid
Insurance covered - Pharmacy cost
Insurance covered - Medical cost
Out of pocket expenditure - Pharmacy Cost
Out of pocket expenditure - Medical Cost

Figure 14. Annual Trends in Disease-Specific Healthcare Expenditure (Per Member Per Month Costs) among Opioid Overdose Patients (2016-2019).

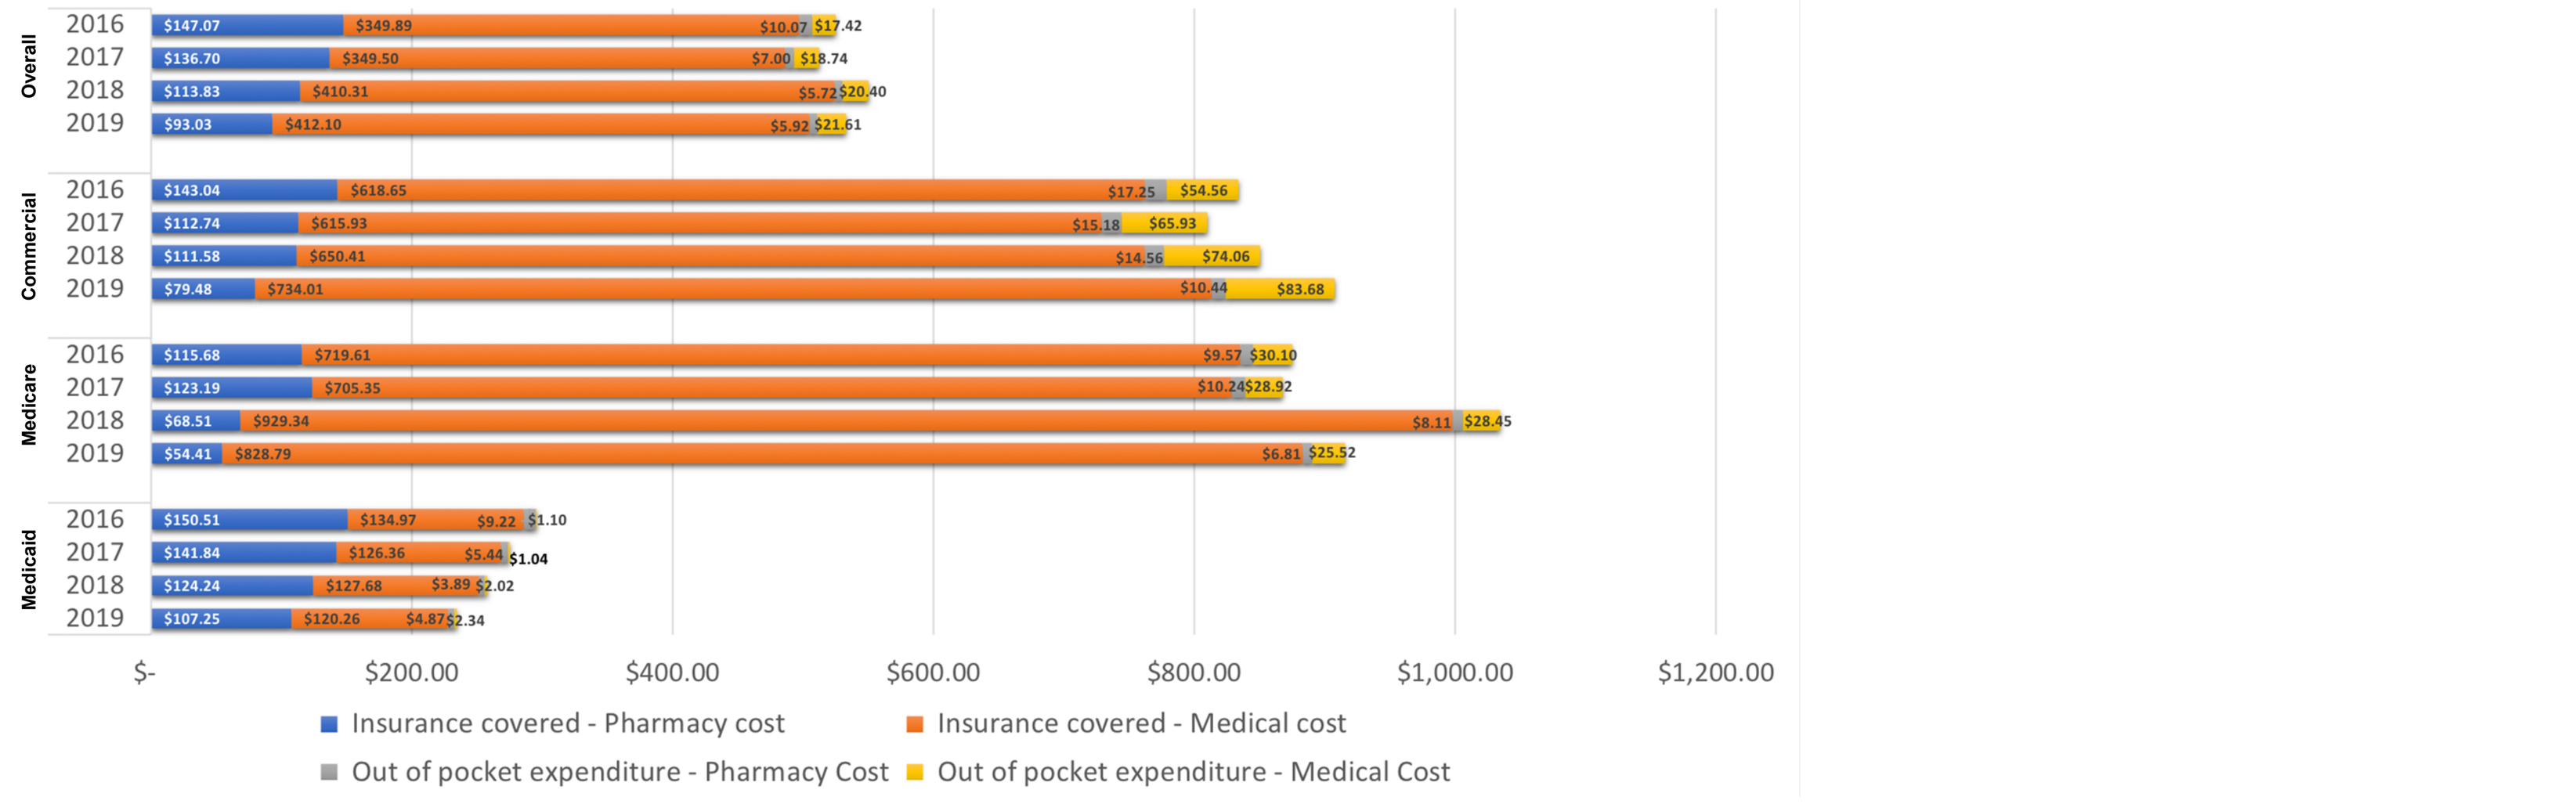
Overall
Commercial
Medicare
Medicaid
Insurance covered - Pharmacy cost
Insurance covered - Medical cost
Out of pocket expenditure - Pharmacy Cost
Out of pocket expenditure - Medical Cost

Key Findings

Characteristics of Opioid Overdose Patients

- The overall number of OOP did not drastically change during the study. 20,694 OOP were identified in 2016, followed by 22,864 in 2017, 20,351 in 2018, and 19,495 in 2019. The majority of OOP were covered by Medicaid across all years, representing 57.85% of OOP in 2016 and 60.37% in 2019. Medicare patients accounted for 23.14% of OOP in 2016, rising slightly to 25.79% in 2019. OOP rates among commercially insured patients changed the most over the duration of the study, decreasing from 19.01% in 2016 to 13.84% in 2019.
- The mean age of the OOP cohort increased from 45.70 in 2016 to 48.80 in 2019. The percentage of OOP that were ≥50 years of age increased by 8.63% between 2016 and 2019, and OOP aged 65-84 years increased by 31.33%. The largest increase in ≥50-year-old OOP between 2016 to 2019 was among Medicaid beneficiaries (5.81%) followed by Medicare (3.04%) and those with commercial insurance (0.72%). The percentage increase of OOP that were 65-84 years old between 2016 to 2019 was highest among individuals with Medicare (7.32%), followed by those with Medicaid (3.10%) and commercial insurance (1.04%).
- The overall number of OOP less than 50 years of age decreased from 59.70% in 2016 to 53.55% in 2019. Three-quarters (75.51%) of OOP with commercial insurance were <50 years of age in 2019, followed by 67.69% with Medicaid and less than one-tenth (8.69%) with Medicare.
- The percentage of OOP that were Caucasian declined by 11.05% during the study period, from 65.97% in 2016 to 54.92% in 2019. The largest decline was observed among Medicare beneficiaries (14.17%), followed by individuals with commercial insurance (13.40%) and Medicaid (8.51%).
- More than half of OOP were male (51.56%), rising to 53.18% in 2019. Between 2016-2019, the largest increase in male OOP was observed among individuals with commercial insurance (4.08%), followed by Medicaid (2.45%) and Medicare (0.21%) beneficiaries. In 2019, OOP with Medicare were predominantly female (59.74%), while males represented the majority of OOP covered by commercial insurance (60.50%) and Medicaid (57.01%).
- States with the highest distribution of OOP in 2019 were Ohio (9.37%), New Jersey (7.04%), New York (6.72%), Florida (6.50%), and Arizona (5.87%). Commercially insured OOP mostly resided in Florida (10.45%), Texas (6.85%), New York (6.26%), California (5.48%), and Ohio (5.11%). OOP with Medicare were mostly from Texas (11.38%), Florida (9.71%), Georgia (7.98%), California (6.21%), and North Carolina (6.01%). States with the most Medicaid OOP included Ohio (13.37%), New Jersey (10.09%), Pennsylvania (8.50%), New York (8.37%), and Arizona (7.21%).

Comorbidities among OOP

- The percentage of OOP with at least one comorbidity increased from 43.24% in 2016 to 46.66% in 2019. In 2019, Medicare OOP had the highest prevalence of comorbidity at 75.69%, followed by Medicaid OOP (38.41%) and commercial patients (28.57%). Medicare OOP also had more severe comorbid conditions in 2019, with an average Charlson Comorbidity Index (CCI) score of 2.37. Medicaid and commercial OOP had lower average CCI scores of 0.91 and 0.66, respectively.
- Between 2016-2019, the most common comorbidities among all OOP included chronic pulmonary disease (19.61%-21.14%), diabetes with chronic complications (10.71-11.30%), congestive heart failure (8.16%-10.12%), mild liver disease (7.16-8.64%), and renal disease (5.59%-6.52%).
- Between 2016-2019, the most common AHRQ1 comorbidity among OOP was mental illness for commercial (73.29%-76.53%) and Medicaid (74.67%-78.83%) patients, whereas diseases of the circulatory system were the most common AHRQ1 comorbidity among Medicare OOP patients (83.27%-85.42%).
- Between 2016-2019, substance-related disorders were the most common AHRQ2 and AHRQ3 comorbidities among commercial (AHRQ2: 41.90% - 44.77%; AHRQ3: 41.90%-44.77%) and Medicaid (AHRQ2: 48.05%-56.96%; AHRQ3: 48.05%-56.96%) OOP. The most common AHRQ2 comorbidities among Medicare OOP were spondylosis, intervertebral disc disorders, and other back problems (61.03%-65.11%). Diseases of the heart were the most common AHRQ3 comorbidities among Medicare OOP (67.96%-70.33%).

Healthcare Costs

- The overall per member per month costs (PMPMC) for OOP in 2019 was $4,310.29, compared with PMPMC of $3,810.37 in 2016, amounting to an increase of 13.12%. Between 2016 to 2019, PMPMC for OOP increased by 18.20% among individuals with Medicare, followed by a 1.54% increase among those with commercial insurance. By contrast, PMPMC for OOP slightly declined (0.66%) among those with Medicaid.
- In 2019, insurance-covered medical costs were $3,541.46, contributing to 82.16% of overall healthcare cost for OOP. Insurance-covered pharmacy costs, out-of-pocket medical costs, and out-of-pocket pharmacy costs averaged $559.84, $154.77, and $54.21, respectively.
- In 2019, out-of-pocket expenditure for OOP was highest among those with commercial insurance ($443.72), followed by Medicare ($298.79) and Medicaid ($94.25) beneficiaries. Between 2016 to 2019 the out-of-pocket expenditure increased by 50.11% for OOP enrolled in Medicaid plans and 9.73% for OOP with commercial insurance. By contrast, out-of-pocket expenditure declined by 11.05% for OOP enrolled in Medicare plans. Insurance covered expenses increased by 19.81% and 0.87% between 2016 to 2019 for OOP with Medicare and commercial insurance, respectively, while it decreased by 2.12% for OOP with Medicaid.
- In 2019, the disease-specific costs for OOP (the costs associated specifically with the treatment of opioid poisoning) were highest for Medicare patients ($915.54), followed by commercial insurance ($907.62) and Medicaid ($234.71) patients. Out-of-pocket expenses were highest for commercial insurance ($94.13), followed by Medicare ($32.33) and Medicaid ($7.20). Insurance-covered expenses for pharmacy and medical costs were highest for Medicare ($883.21), followed by commercial plans ($813.49) and Medicaid ($227.51).

Limitations

- Undetected coding errors made by providers may have resulted in incomplete, inaccurate, or missing claims data that led to the misidentification of OOP.
- Opioid overdose cases treated outside of healthcare provider settings (no filed claims) were not captured in our dataset.
- Patient inclusion criteria for this claims analysis included any opioid overdose diagnosis. Consequently, this study did not examine whether opioid drugs involved in overdoses were obtained illicitly or by prescription.
- Our analysis was restricted to a defined study window with a minimum continuous enrollment of 30 days pre- and post-index event (first date of opioid overdose claim); therefore, our study did not address overdose-induced deaths occurring at the index event.
- Limitations of study design must be considered when reviewing the findings from this study.

Conclusions

- The findings of this study add to the existing body of literature by providing previously unavailable data on the characteristics and expenditure of opioid overdose patients broken down by payor and year.
- Opioid overdoses resulted in significant economic burden to patients and payors in the healthcare industry. In 2019, Medicare beneficiaries had the highest total PMPMC across four categories (insured medical costs, insured pharmacy costs, out-of-pocket medical costs, and out-of-pocket pharmacy costs) as well as the highest disease-specific insured medical costs. Commercial insurance beneficiaries had the highest disease-specific PMPMC for insured pharmacy costs, out-of-pocket medical costs, and out-of-pocket pharmacy costs.
- This study contributes important data on opioid overdose patient trends, costs, and comorbidities and may serve as an important resource for public health policymakers, researchers, and payors.

Future Research

- Further analysis is needed to evaluate the association between baseline comorbidities and risk of opioid overdose.
- Future studies should investigate the potential benefits of naloxone and determine if naloxone prescribing helps to reduce healthcare costs among OOP.
- Additional research is needed to investigate the increasing number of opioid overdoses among individuals aged 50 years or older, as this observed trend may result in significant burden on the healthcare system.

Acknowledgements

The authors of this presentation would like to thank ScioScientific for their contributions to this poster.

References

1. Substance Abuse and Mental Health Services Administration. (2021). Key substance use and mental health indicators in the United States: Results from the 2020 National Survey on Drug Use and Health (HHS Publication No. PEP21-07-01-003, NSDUH Series H-56). Rockville, MD: Center for Behavioral Health Statistics and Quality, Substance Abuse and Mental Health Services Administration.
2. National Institute on Drug Abuse. Trends and Statistics: Overdose Death Rates. https://nida.nih.gov/drug-topics/trends-statistics/overdose-death-rates; retrieved January 27, 2022.
3. Soares WE 3rd, Melnick ER, Nath B, D'Onofrio G, Paek H, Skains RM, Walter LA, Casey MF, Napoli A, Hoppe JA, Jeffery MM. Emergency Department Visits for Nonfatal Opioid Overdose During the COVID-19 Pandemic Across Six US Health Care Systems. *Ann Emerg Med*. 2022 Feb;79(2):158-167.
4. Premier Inc. "Opioid Overdoses Costing U.S. Hospitals an Estimated $11 Billion Annually," news release, Jan. 3, 2019. https://www.premierinc.com/newsroom/press-releases/opioid-overdoses-costing-us-hospitals-an-estimated-11-billion-annually; retrieved February 8, 2022.
5. Florence C, Luo F, Rice K. The economic burden of opioid use disorder and fatal opioid overdose in the United States, 2017. *Drug Alcohol Depend*. 2021 Jan 1;218:108350.
6. Centers for Disease Control and Prevention. Life-Saving Naloxone from Pharmacies. https://www.cdc.gov/vitalsigns/naloxone/index.html; retrieved February 21, 2022.
7. Optum's de-identified Clinformatics® Data Mart Database (2007-2021).