EE129. A COST-EFFECTIVENESS ANALYSIS FOR TERLIPRESSIN IN TREATING BLEEDING OESOPHAGEAL VARICES PATIENTS IN THE PHILIPPINES

Yasay E,[1] Lai A,[2,3] Irmawan AD,[2] Panlilio MT,[4] Ong J,[5] Wu E,[6] Kim J,[6] Wang B,[6]

[1] Providence Hospital, The Philippines, [2] Ferring Pharmaceuticals, [3] National Taiwan University, Taiwan, [4] National Kidney and Transplant Institute, The Medical City, The Philippines, [5]Philippines General Hospital, The Philippines, [6]Real Chemistry Consulting, NY, USA,

Objective

Bleeding Oesophageal Varices (BOV) is a life-threatening condition that affects approximately 50% of cirrhotic patients. Vasoactive agents such as octreotide, somatostatin, and terlipressin are mainstays in the management BOV, with efficacy in attaining hemostasis in about 75% and 67% of patients at 48 hours and at 5 days, respectively. We conducted a cost-effectiveness analysis of terlipressin in the treatment of BOV from the payer's perspective in the Philippine setting.

Method

- **Model type:** a discrete event simulation with five health states (Bleed, No Bleed, No Bleed post transjugular intrahepatic portosystemic shunt (TIPS), Post salvage Surgery and Death) and six events (bleed stopped, bleed >14 days, re-bleed, TIPS performed, salvage surgery and death) **(Figure 1)**
- **Time horizon:** one year
- **Simulation cohort size:** 1000
- **Comparators:** octreotide, somatostatin, terlipressin and placebo.
- **Clinical inputs:**
 - Baseline risk curves were fitted to approximate baseline survival, re-bleeding rates, and failure to control bleeding for each treatment based on published data. General Weibull was chosen as the best fit.
 - Relative risks of mortality, re-bleeding rate and failure to control bleeding were obtained from the Cochrane meta-analysis.[1-2] It was assumed that somatostatin and octreotide have identical relative risks as trials for both were not pooled separately. Relative risks were summarized in **Table 1.**
 - A baseline health state utility for non-bleeding BOV patients of 0.75 was based on literature.[3-4] A disutility of 25% from baseline was applied for each bleeding episode according to clinical expert's opinions. The utility related to TIPS and salvage surgery were 0.25 and 0.5. **(Table 1)**
- **Economic inputs:** the unit costs of health services and corresponding utilization were informed by local cost databases and the clinical experts in the Philippines.[5-7] **(Table 2)**
- **Model outcomes:** total costs in the Philippines pesos (₱), quality-adjusted life-year (QALYs) and incremental cost-effectiveness ratio (ICER) **(Table 3)**

Figure 1. Model Structure

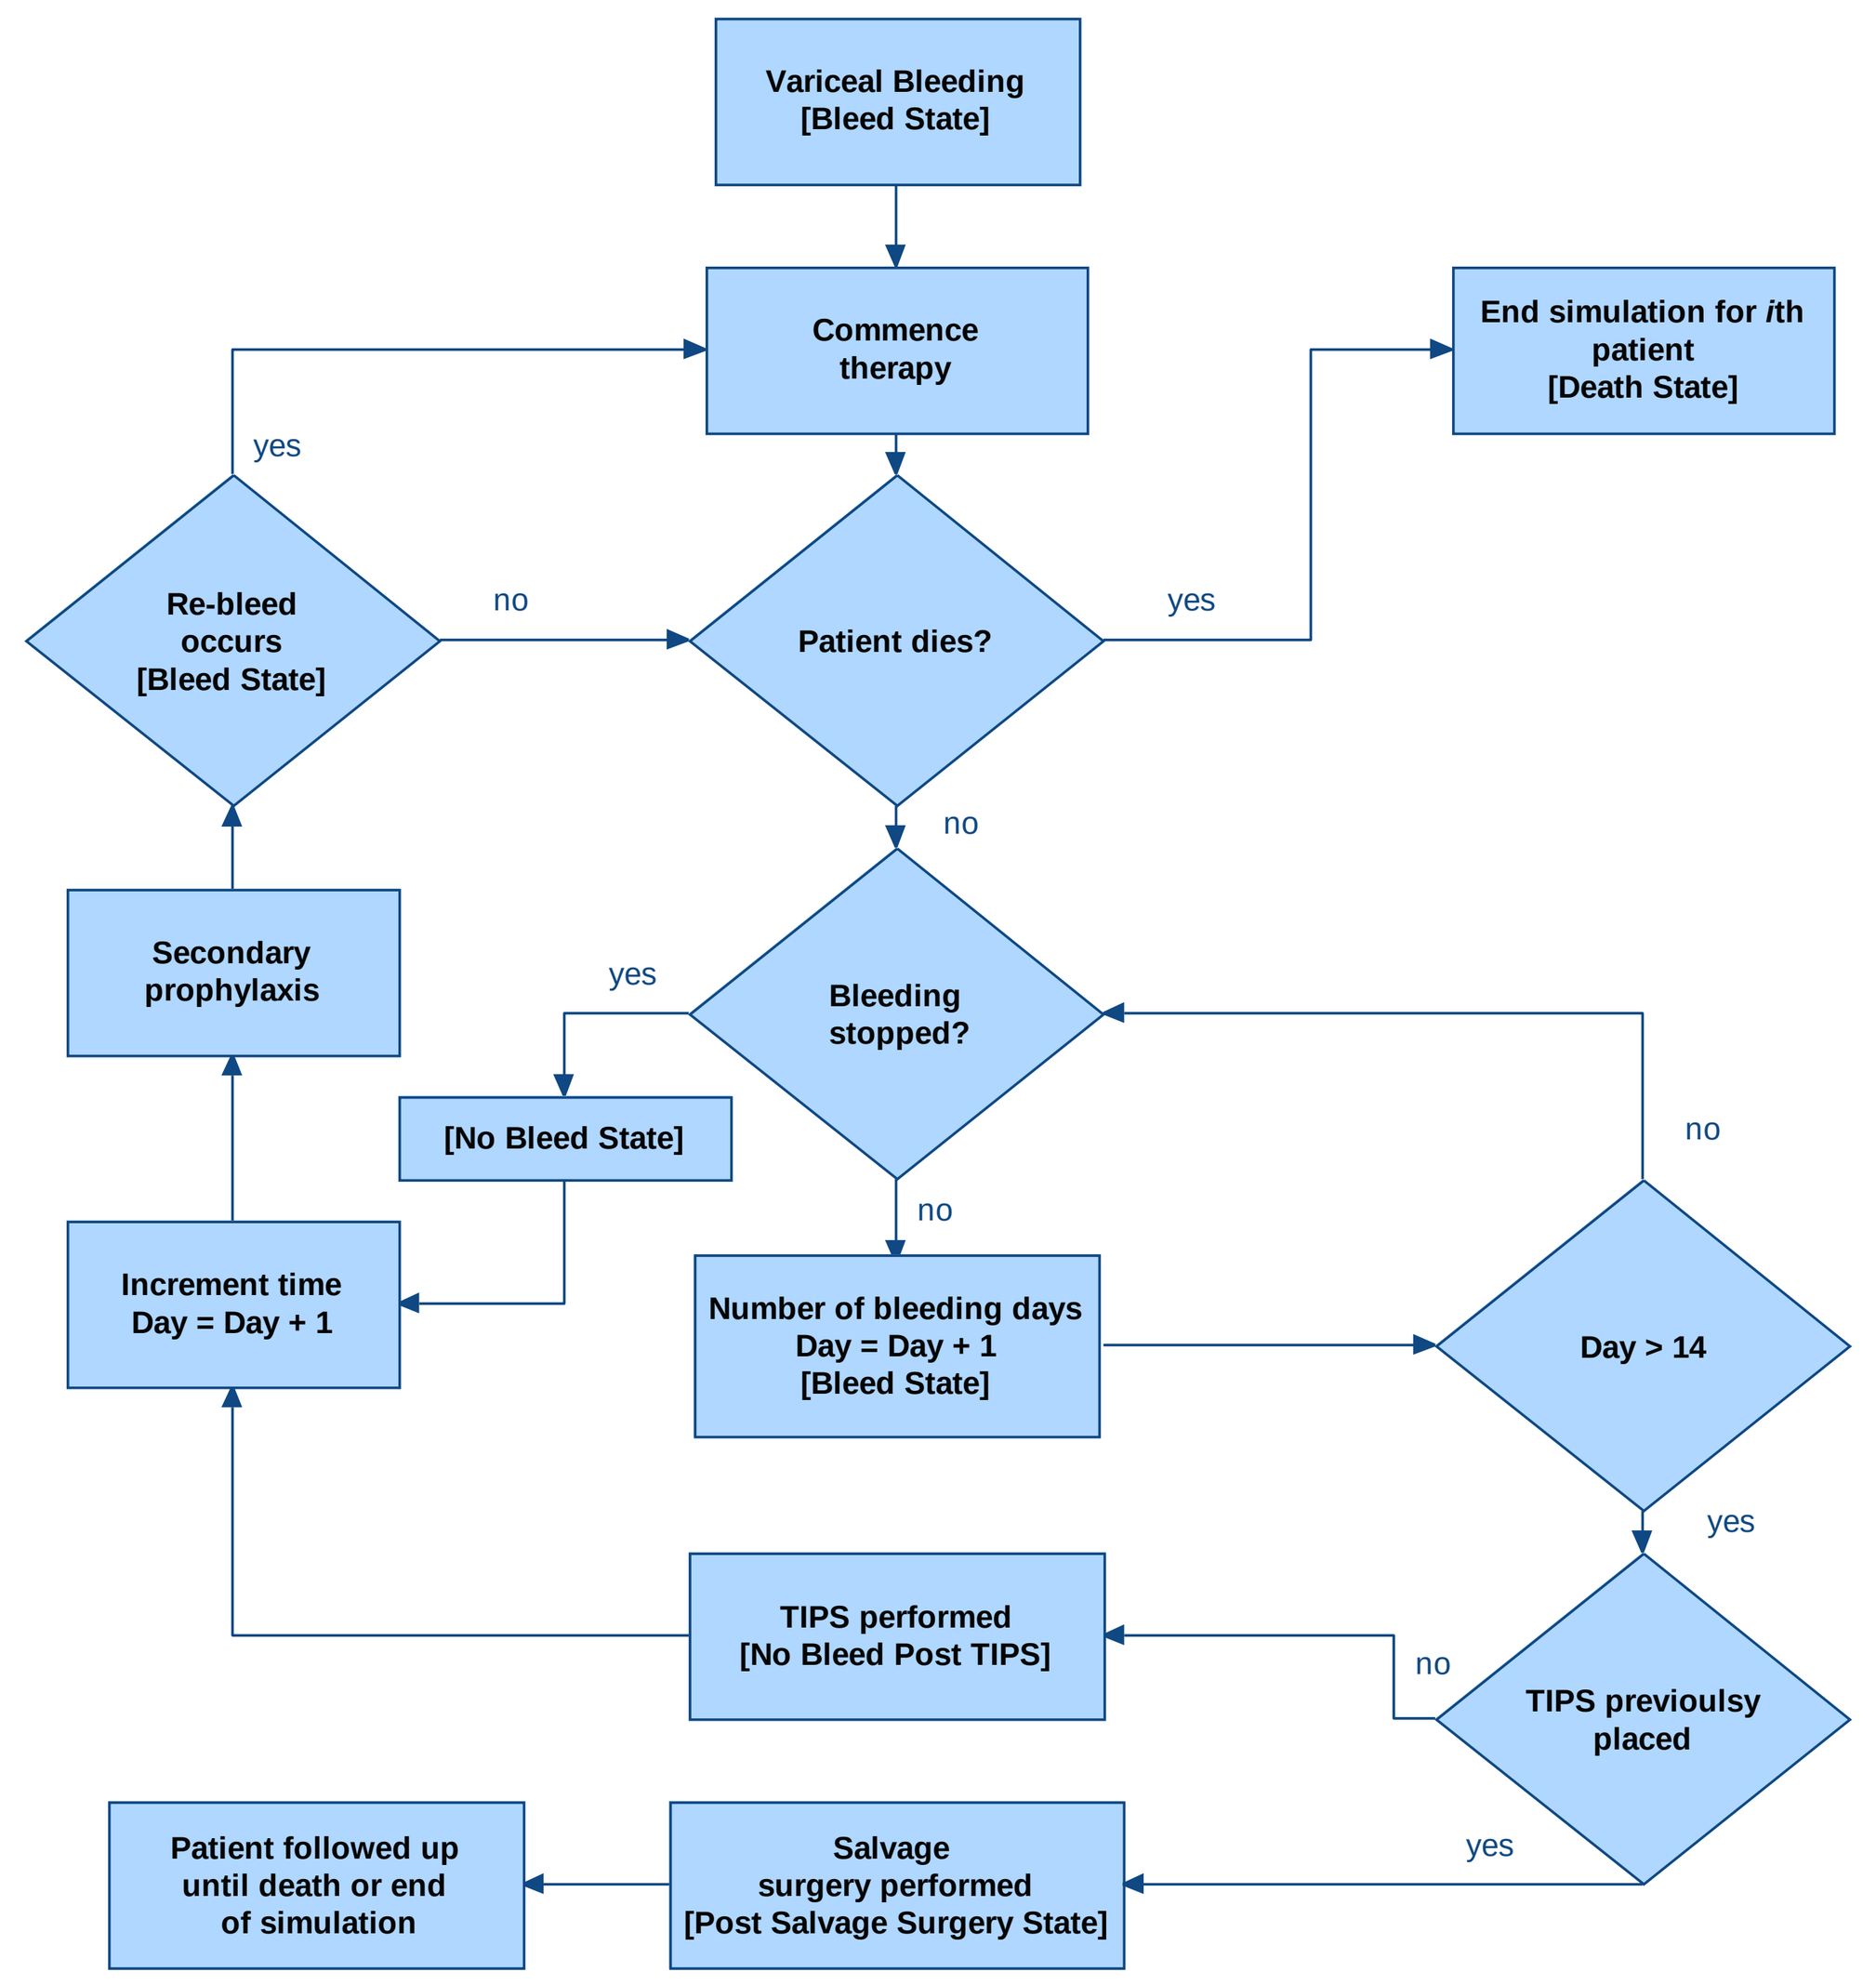

Variceal Bleeding
[Bleed State]
Commence therapy
End simulation for *i*th patient
[Death State]
yes
Re-bleed occurs
[Bleed State]
no
Patient dies?
yes
no
Secondary prophylaxis
yes
Bleeding stopped?
no
[No Bleed State]
no
Increment time
Day = Day + 1
Number of bleeding days
Day = Day + 1
[Bleed State]
Day > 14
yes
TIPS performed
[No Bleed Post TIPS]
no
TIPS previoulsy placed
yes
Patient followed up until death or end of simulation
Salvage surgery performed
[Post Salvage Surgery State]

Table 1. Clinical Efficacy

Parameters	Value
Relative Risks vs. Placebo [1-2]	
Bleeding (octreotide)	0.65
Bleeding (somatostatin)	0.65
Bleeding (terlipressin)	0.45
Re-bleeding (octreotide)	0.82
Re-bleeding (somatostatin)	0.82
Re-bleeding (terlipressin)	0.93
Death (octreotide)	0.96
Death (somatostatin)	0.96
Death (terlipressin)	0.61
Utilities[3-4]	
Multiplier for cirrhosis	0.75
Bleeding episodes	0.25
Post-salvage surgery	0.5
Post-TIPS	0.25

Table 2. Economic Inputs

Parameters	Value
Hospitalization during the bleeding[5]	
Non-ICU	₱ 500
Intensive Care	₱1,500
Rescue Surgery[5]	
TIPS	₱ 200,000
Salvage Surgery	₱ 200,000
Secondary prophylaxis[5]	
Band Ligation	₱ 7,000
Physician Consultation	₱ 500
Drug Costs (per vial/per tab) [6-7]	
terlipressin	₱ 1840
Octreotide	₱ 4491
Somatostatin)	₱ 5109
Propranolol	₱ 35.2
Urso/Antibiotic	₱ 50

Results

- The results showed that treatment with terlipressin was associated with an additional ₱395 per patient (compared with somatostatin), a cost-saving of ₱1,563 per patient (compared with octreotide), and an additional of ₱20,236 per patient (compared with placebo). Moreover, the clinical benefit for a patient with BOV over one year manifested as an increase in an incremental QALY gain for terlipressin of 0.0932 (compared with somatostatin and octreotide) and 0.0934 (compared with placebo).
- Terlipressin was cost-effective when compared to somatostatin (₱4,241), octreotide (-₱16,767) and placebo (₱216,555 or approximately 4,336 USD per QALY). In the Philippines, one gross domestic product (GDP) per capita is approximately 3,330.36 USD.[8] The ICERs are within the WHO-recommended recommended cost-effectiveness thresholds of 1 to 3 times the GDP per capita.[9]
- One-way deterministic sensitivity analysis indicated that our data was robust with ICER results driven by drug costs and length of treatment.

Table 3. Cost-Effectiveness Analysis Results

	Terlipressin	Somatostatin	Octreotide	Placebo
Costs	₱44,115	₱43,719	₱45,678	₱23,878
QALYs	0.58	0.48	0.48	0.48
Differences in Costs (vs. Terlipressin)		₱395	-₱1,563	₱20,236
Differences in QALYs		0.0932	0.0932	0.0934
ICER		₱4,241	-₱16,767 (Dominates)	₱216,555

Conclusion

Vasoactive drugs are cost effective in managing BOV. Among the three vasoactive agents studied, terlipressin was the most cost-effective pharmacologic intervention in treating BOV in the Philippines from a public payer's perspective.

References

1. Ioannou G et al. Cochrane Database Syst Rev. 2003
2. Gotzsche et al. Cochrane Database Syst Rev. 2005
3. Rubenstein et al. Am J Gastroenterol 2004;99:1274
4. Younossi et al. Am J Gastroenterol 2001;96:579
5. Local Clinical Expert Opinion
6. Drug Price Index — Department of Health
7. The Filipino Doctor
8. Statista 2021
9. WHO 2021

Disclosures: This study was funded by Ferring Pharmaceuticals

ISPOR Annual USA Congress, May 15th – May 18th, Washington, DC, USA

FERRING PHARMACEUTICALS