Twelve-Month Inpatient Mortality and Economic Burden of *Clostridioides difficile* Infections in the US: 2016-2019

Tillotson G[1], Le J[2], Lodise TP[3], Bochan M[4], Ng S[5], Poole CD[6], Guo A[5], Feuerstadt P[7]

[1]GST Micro LLC, North, VA, USA; [2]Xcenda, Carrollton, TX, USA; [3]Albany College of Pharmacy and Health Sciences, Watervliet, NY, USA; [4]Ascension St. Vincent Indianapolis, Carmel, IN, USA; [5]Ferring Pharmaceuticals, Inc., Parsippany, NJ, USA; [6]Xcenda, London, UK; [7]Yale New Haven Hospital/PACT Gastroenterology Center, Westport, CT, USA

FERRING PHARMACEUTICALS

BACKGROUND

- *Clostridioides difficile* infections (CDI) are the most common healthcare–associated infections in the United States (US), with around 420,000 new cases per annum[1]
- CDI is associated with serious acute complications, including sepsis, surgical bowel resection, death, and recurrence episodes[2]
- Acute care facility costs are the main drivers of the overall economic burden of CDI estimated at $4.8 billion in 2015[3,4]
- Despite detailed knowledge of the overall epidemiology and burden of CDI, limited data are available on its effect on US hospitals

OBJECTIVES

- To determine the 12-month frequency of all-cause and CDI-related rehospitalization following index hospitalization with a CDI diagnosis
- To quantify complication rates, inpatient mortality, cost of care, and expected reimbursement for each all-cause and CDI-related hospitalization during the 12-month follow-up period

METHODS

Study Design (Figure 1)

- A retrospective cohort study using hospitalization data from the Premier Healthcare Database[5] (PHD) between January 1, 2016, and September 30, 2019 was performed
- Hospitalizations with a primary or secondary discharge diagnosis of CDI were considered CDI-related
- The index event was the patient's first CDI-related hospitalization between January 1, 2017, and September 30, 2018
- Patients were excluded if they had a CDI-related hospitalization in the 12 months prior to their index event
- Patients were followed for 12 months post index or until death
- Demographic characteristics at index included age, gender, race, and primary payer
- The Medicare 5% Standard Analytic File (SAF) was used to estimate reimbursements

Outcomes (12 months post index)

- Primary diagnosis (inferred from first Medicare Severity Diagnosis-Related Group [MS-DRG]), at index, all-cause and CDI-related rehospitalizations
- Rehospitalization rate following index, by all-cause and also CDI-related admission type
- Inpatient mortality (discharge status "expired"), at index, all-cause and CDI-related rehospitalizations
- Length of stay (including intensive care unit [ICU] use, measured in days) by admission type and rehospitalization number
- Clinical complication rates for sepsis (MS-DRGs #870, #871, #872) and bowel surgery (#329, #330, #331, #344, #345, #346)
- Hospital provider costs (US$ 2019) incurred for index, all-cause and CDI-related rehospitalizations
- Estimated reimbursement (calculated in the SAF and applied to each hospitalization in PHD with matching MS-DRG

Analysis

- Descriptive statistics, including means, standard deviations (SD), medians, frequencies, and percentages were used

Figure 1. Study design schema

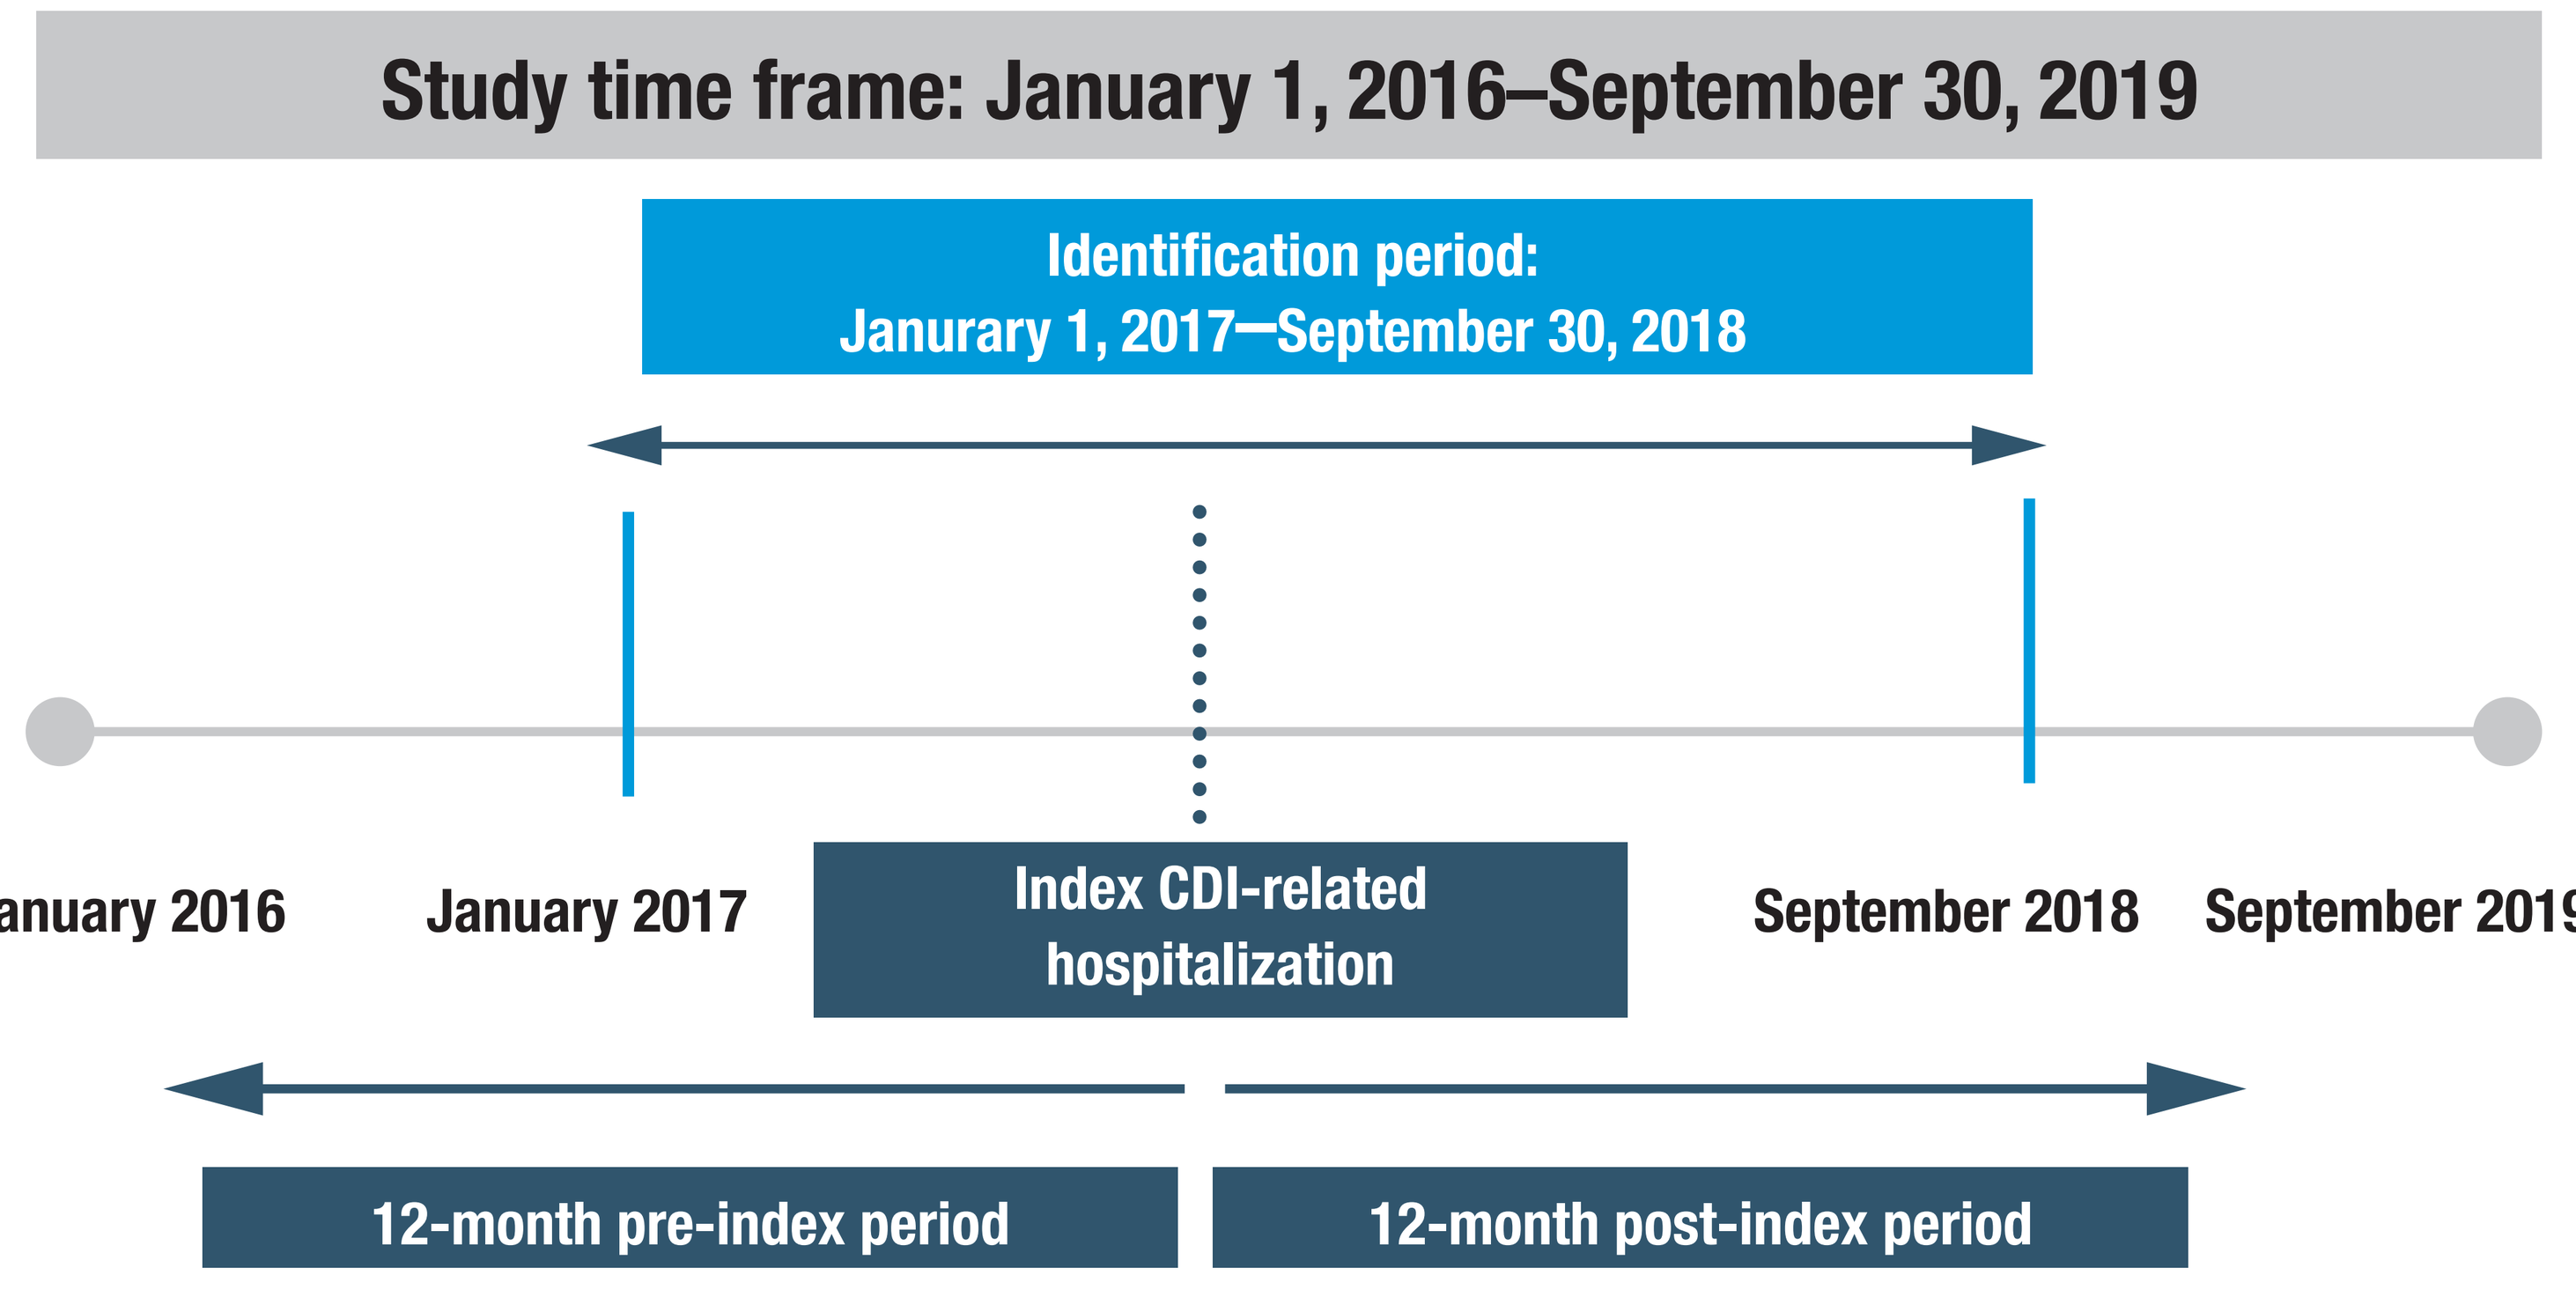
Study time frame: January 1, 2016–September 30, 2019
Identification period:
Janurary 1, 2017—September 30, 2018
January 2016
January 2017
Index CDI-related hospitalization
September 2018
September 2019
12-month pre-index period
12-month post-index period

CDI, *Clostridioides difficile* infection; ICD-10-CM, International Classification of Diseases, 10th Revision, Clinical Modification.
Note: Figure is not drawn to scale and is meant for illustrative purposes only. CDI includes ICD-10-CM A04.7, A04.71, A04.72.

KEY TAKEAWAYS

1 **Inpatient costs, rehospitalization, and mortality for hospitalized patients with CDI are substantial**

2 **For patients with serial readmission, cumulative hospital costs increase significantly as the number of CDI-related hospitalizations increase**

3 **Findings suggest that new treatments and approaches are needed to minimize CDI-related rehospitalizations, deaths, and associated healthcare cost**

Table 1. Top 10 MS-DRGs by admission type and rehospitalization

MS-DRGs	Index	All-cause rehospitalizations					CDI-related rehospitalizations				
		1st	2nd	3rd	4th	≥5th	1st	2nd	3rd	4th	≥5th
372-MJR GI DISORDRS & PERITONEAL INFECT WCC	15.5%	5.3%	3.7%	2.8%	2.2%	2.1%	15.2%	16.6%	16.5%	16.0%	20.1%
871-SEPTICEMIA/SEVR SEPSIS W/ OMV >96HRS WMCC	15.1%	11.3%	11.1%	10.2%	10.1%	8.4%	14.8%	14.4%	12.5%	14.3%	8.3%
371-MJR GI DISORDRS & PERITONEAL INFECT WMCC	8.3%	3.2%	2.4%	2.0%	1.6%	1.7%	9.5%	10.4%	11.6%	11.9%	9.8%
872-SEPTICEMIA/SEVR SEPSS W/ OMV >96HRSW/OMCC	5.9%	3.4%	2.9%	2.4%	2.1%	1.9%	5.6%	5.2%	4.6%	4.2%	5.2%
373-MJR GI DISORDRS&PERITNL INFECT W/OCC/MCC	4.7%	1.4%	0.8%	0.5%	0.3%	0.3%	3.8%	3.4%	2.9%	2.6%	4.3%
853-INFECTIOUS & PARASITIC DIS W OR PX WMCC	3.1%	1.7%	1.8%	1.6%	1.6%	1.6%	2.3%	2.4%	2.0%	1.4%	2.0%
870-SEPTICEMIA OR SEVERE SEPSIS WMV >96 HOUR	1.5%	0.8%	0.7%	0.8%	0.6%	0.8%	1.1%	1.1%	0.9%	1.2%	1.4%
291-HEART FAILURE & SHOCK W MCC	1.4%	3.3%	3.9%	4.5%	4.5%	4.7%	2.2%	2.4%	2.3%	2.4%	2.9%
682-RENAL FAILURE W MCC	1.2%	1.7%	1.7%	1.7%	1.6%	1.6%	1.3%	1.3%	1.2%	0.4%	0.3%
3-ECMO/TRCHWMV>96HR/ PDXEXFCE/MTH&NCKW/MJOR	1.1%	0.1%	0.1%	0.1%	0.1%	0.1%	0.1%	0.1%	0.1%	0.0%	0.0%

CDI, *Clostridioides difficile* infection; MS-DRG, Medicare Severity Diagnosis-Related Group.
Note: The table is sorted in descending order of the MS-DRGs related to the index hospitalization. Gray font indicates that the MS-DRG was not in the top 10 list for that hospitalization.

During the index hospitalization, 22.5% of patients had sepsis

Rehospitalization sepsis rates varied between 13% and 22%

Figure 2. Proportion of patients rehospitalized

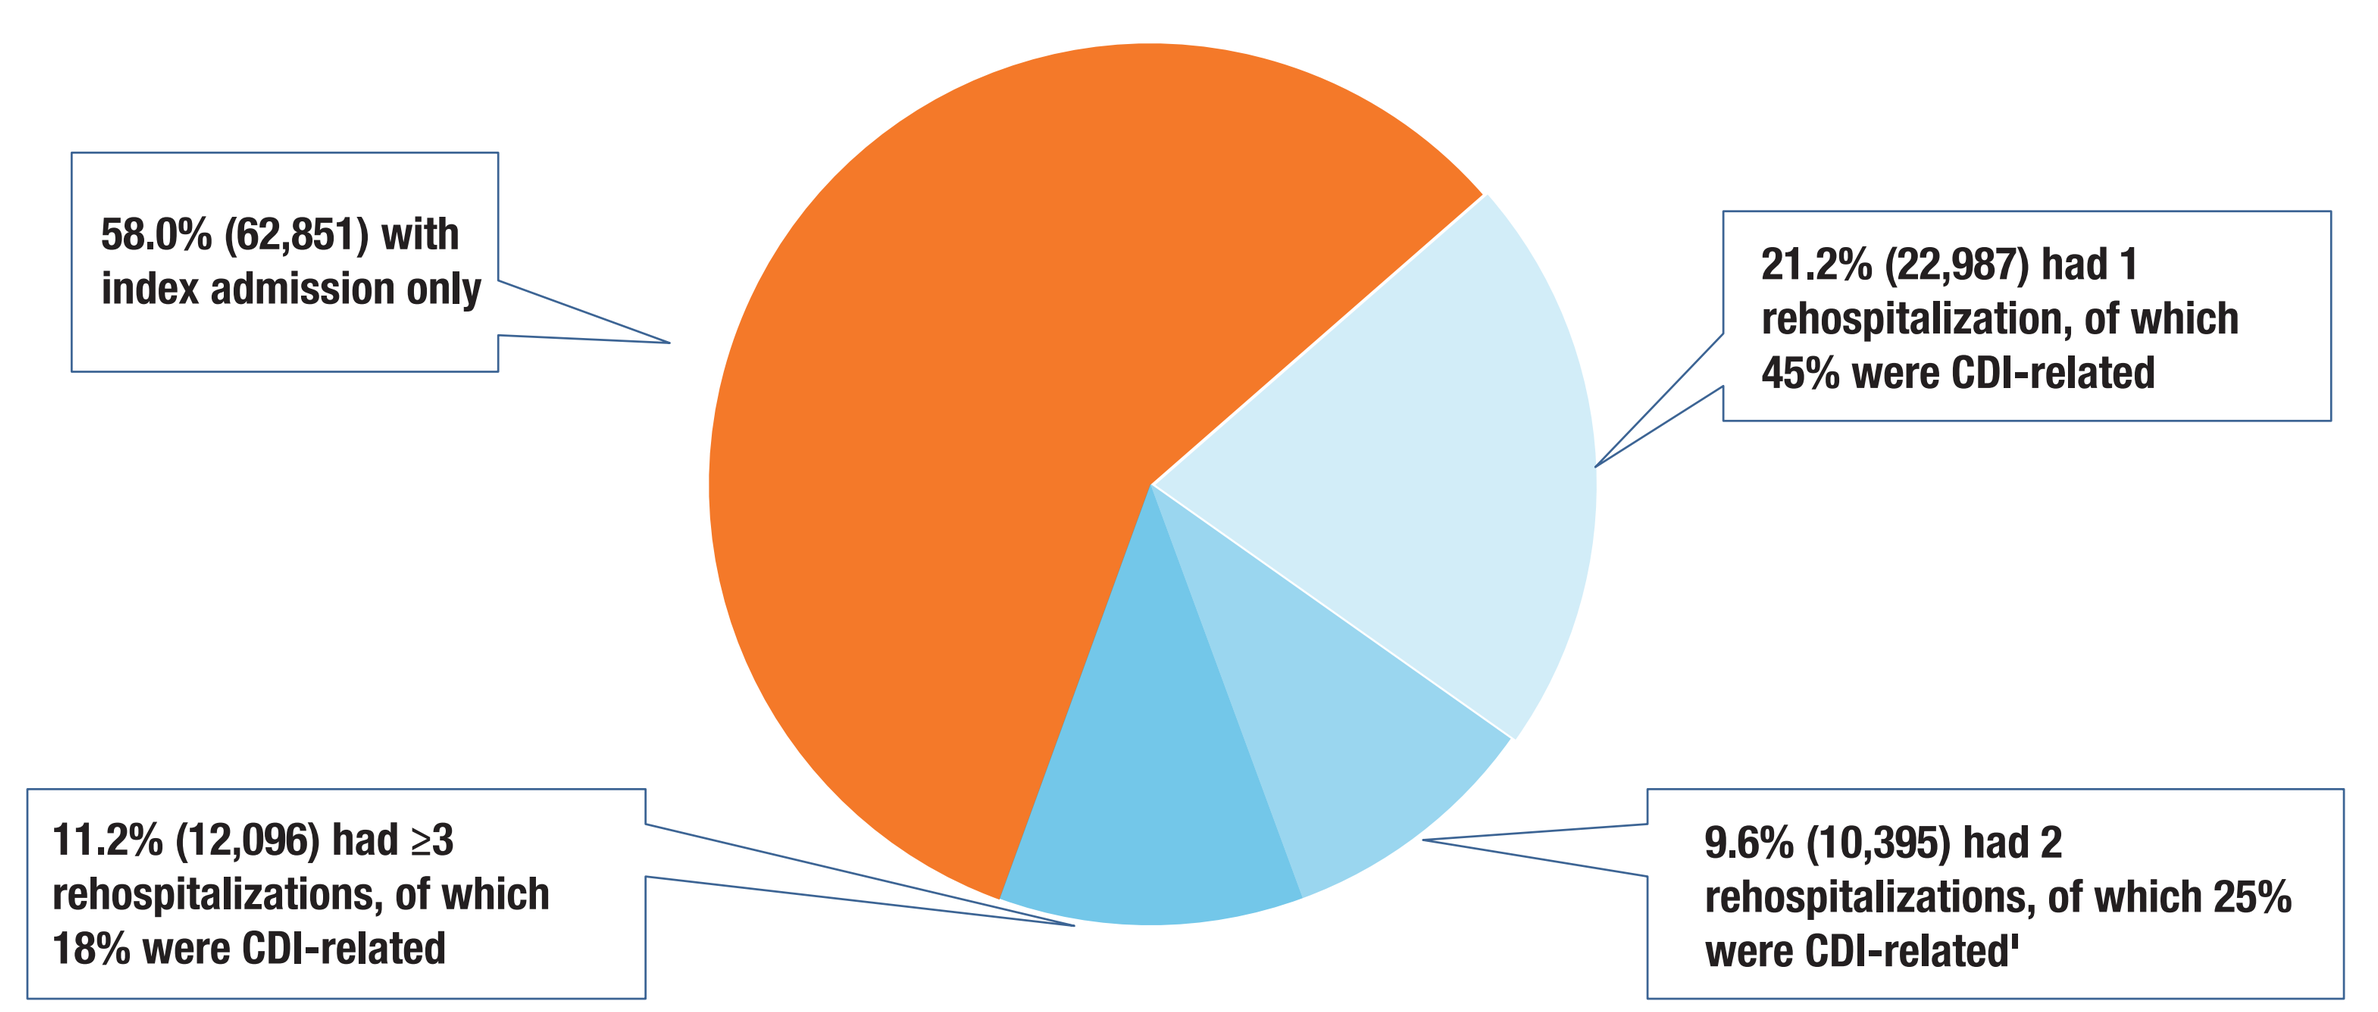
58.0% (62,851) with index admission only
21.2% (22,987) had 1 rehospitalization, of which 45% were CDI-related
9.6% (10,395) had 2 rehospitalizations, of which 25% were CDI-related'
11.2% (12,096) had ≥3 rehospitalizations, of which 18% were CDI-related

Figure 3. Inpatient mortality arising from index hospitalization, all-cause rehospitalization, and CDI-related rehospitalization

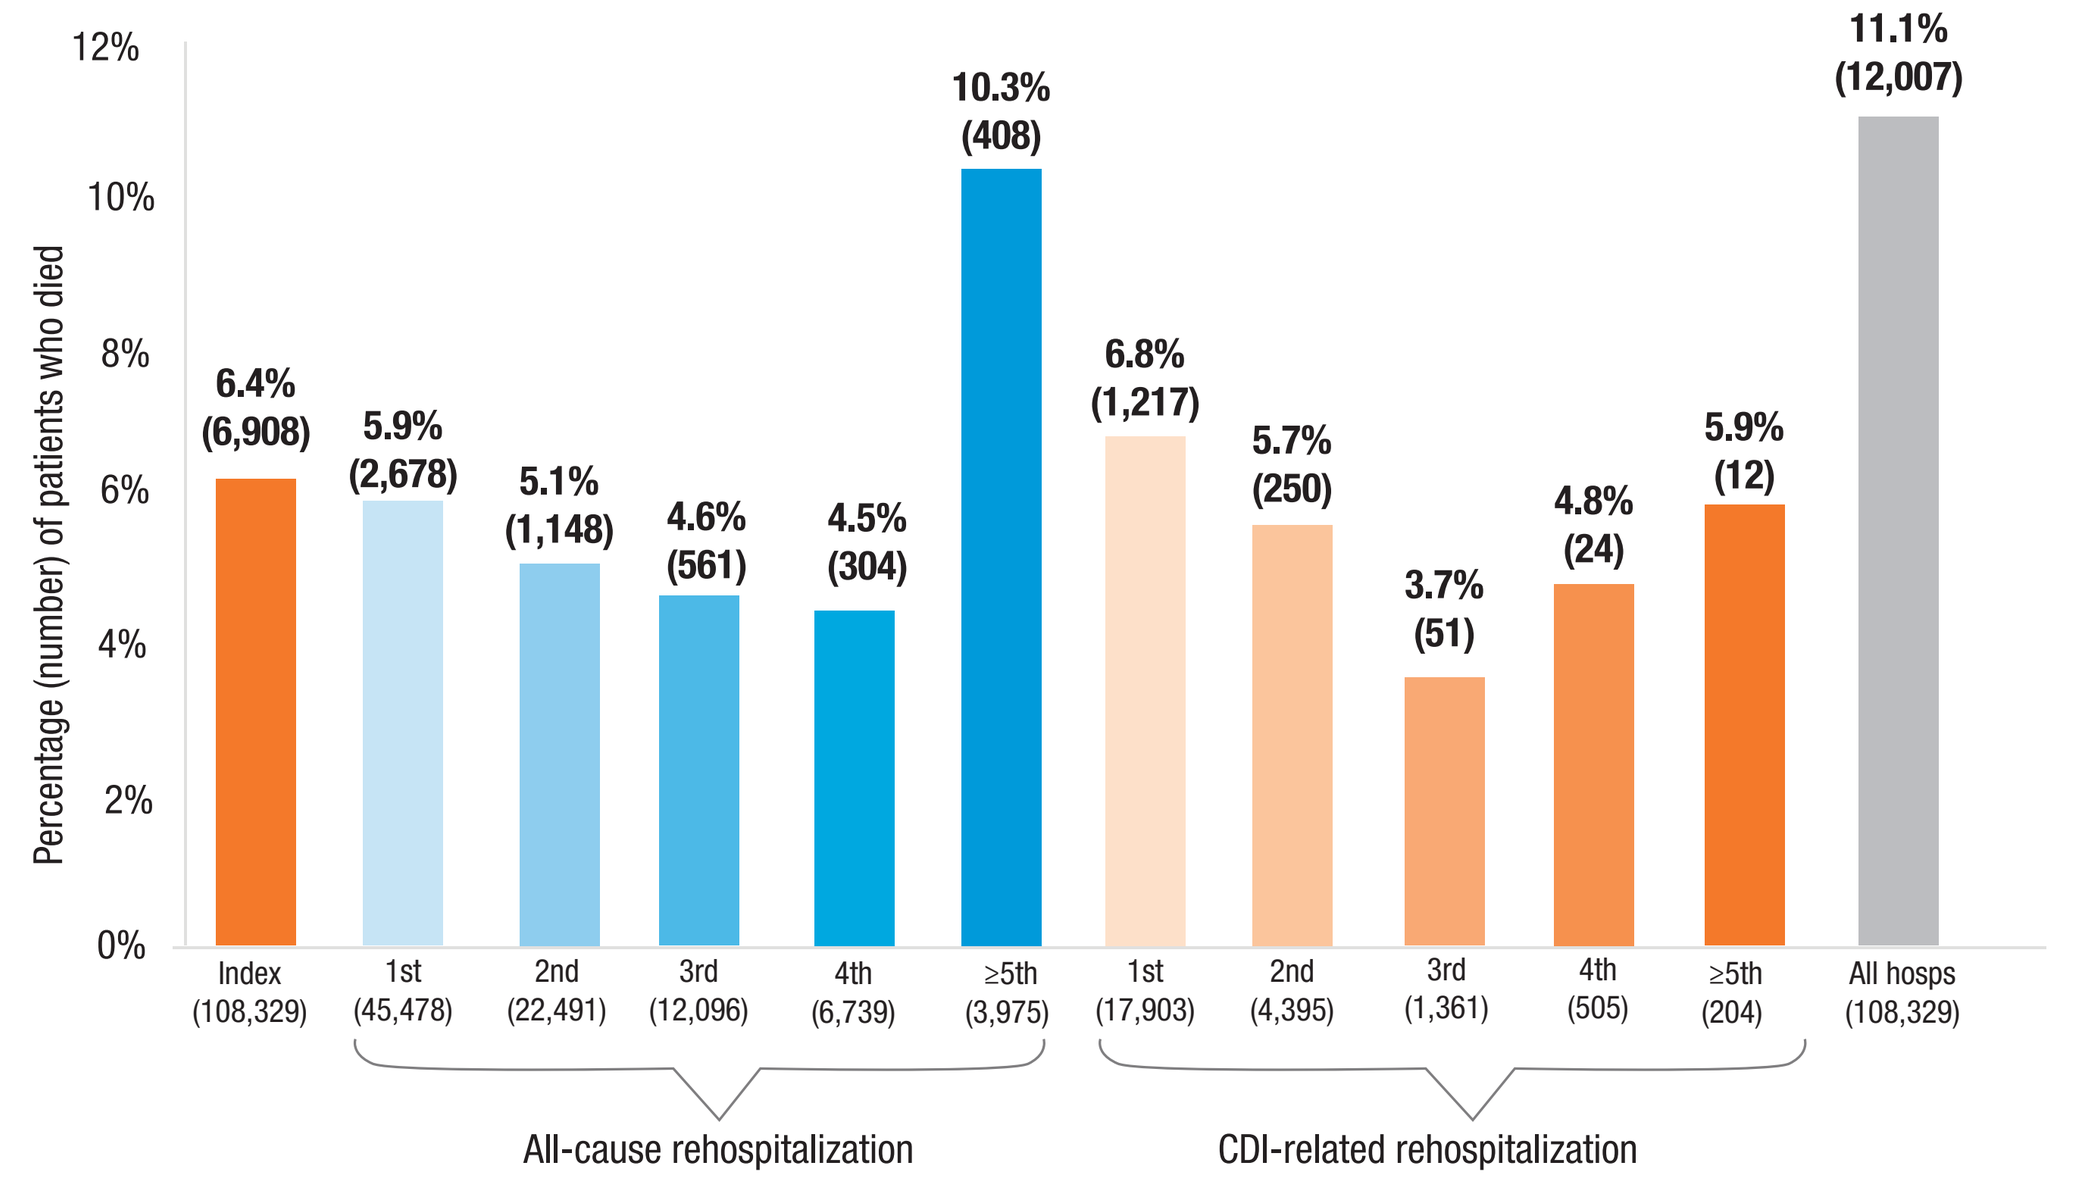
Percentage (number) of patients who died
6.4% (6,908)
5.9% (2,678)
5.1% (1,148)
4.6% (561)
4.5% (304)
10.3% (408)
6.8% (1,217)
5.7% (250)
3.7% (51)
4.8% (24)
5.9% (12)
11.1% (12,007)
Index (108,329)
1st (45,478)
2nd (22,491)
3rd (12,096)
4th (6,739)
≥5th (3,975)
1st (17,903)
2nd (4,395)
3rd (1,361)
4th (505)
≥5th (204)
All hosps (108,329)
All-cause rehospitalization
CDI-related rehospitalization

CDI, *Clostridioides difficile* infection; hosp, hospitalization; rehosp, rehospitalization.

Over 12 months, 11% of patients died during an inpatient stay

Figure 4. Average annual length of stay from index CDI hospitalization and all-cause rehospitalizations

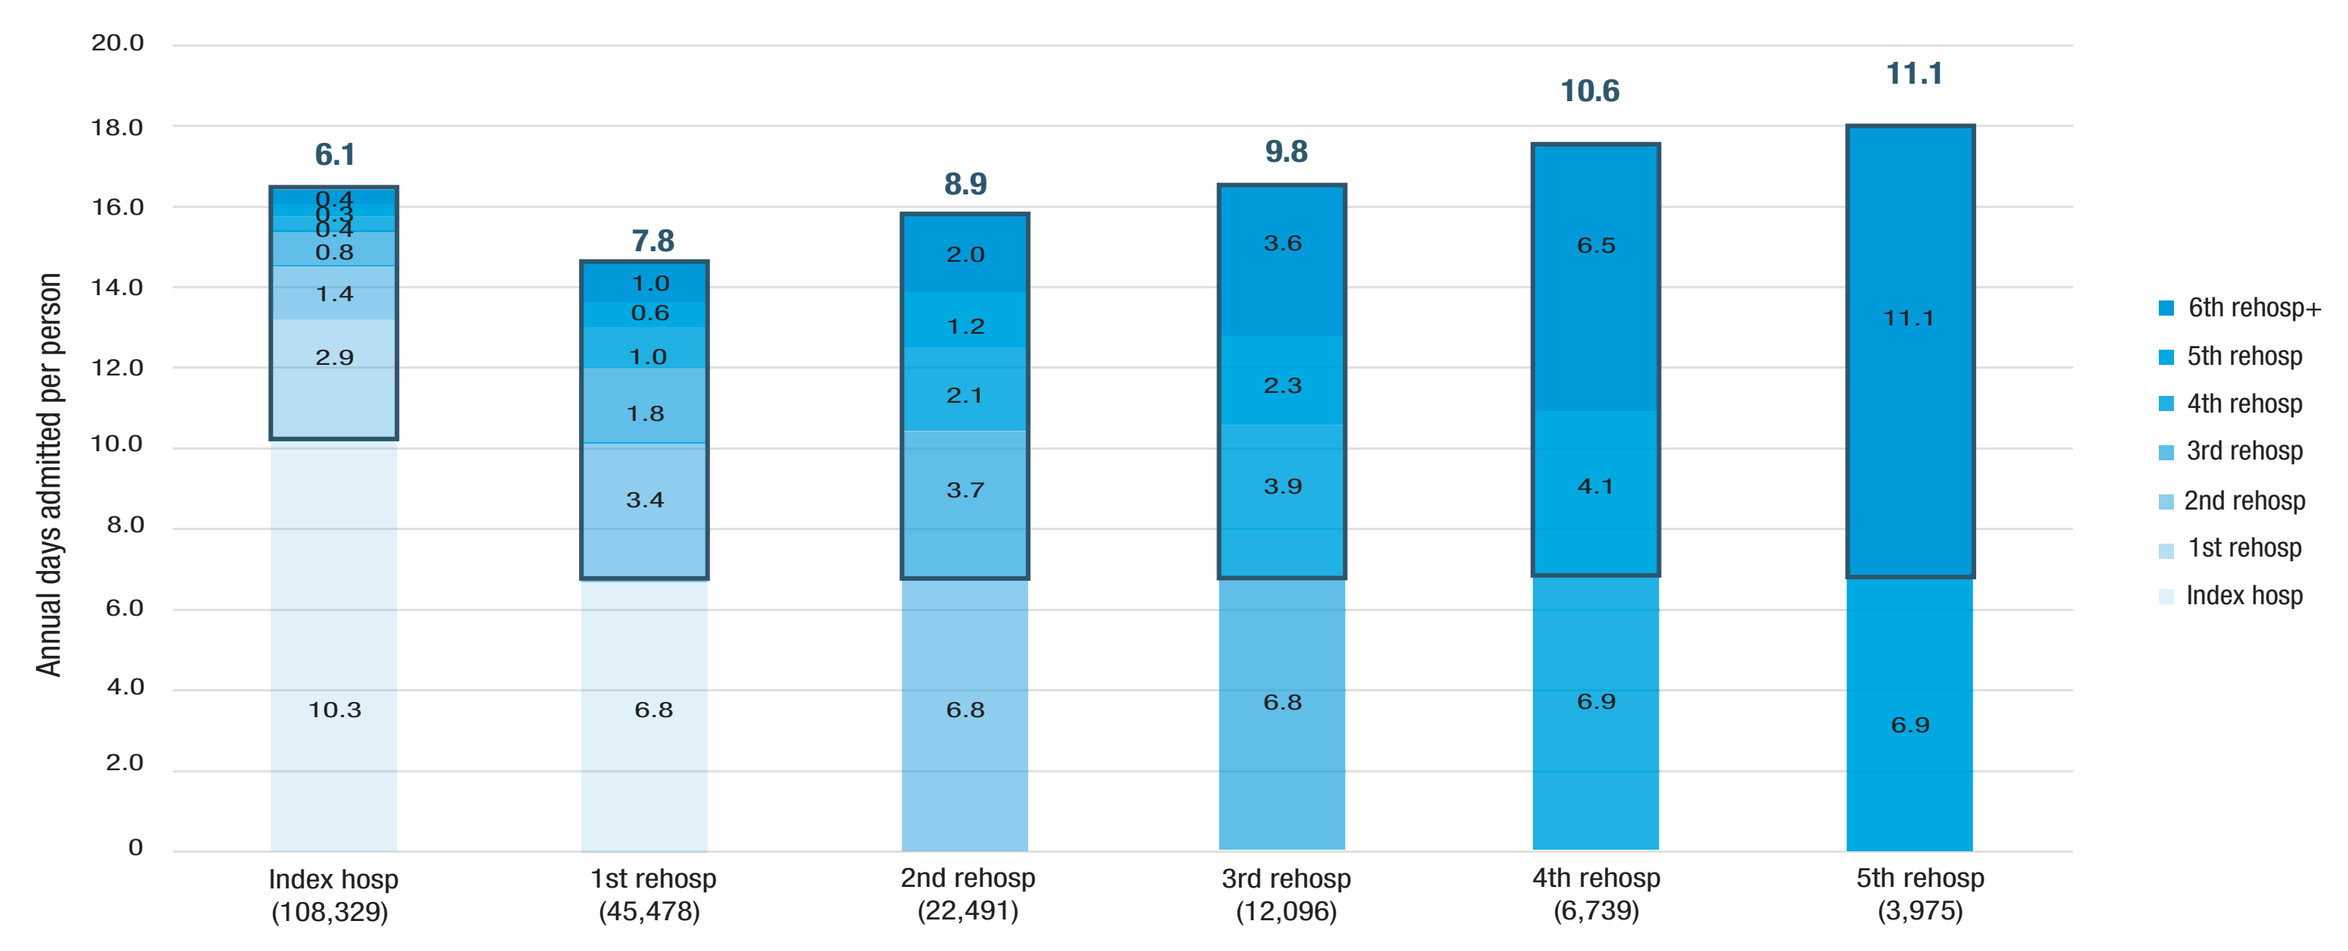
Annual days admitted per person
6.1
7.8
8.9
9.8
10.6
11.1
10.3
6.8
6.8
6.8
6.9
6.9
Index hosp (108,329)
1st rehosp (45,478)
2nd rehosp (22,491)
3rd rehosp (12,096)
4th rehosp (6,739)
5th rehosp (3,975)
6th rehosp+
5th rehosp
4th rehosp
3rd rehosp
2nd rehosp
1st rehosp
Index hosp

hosp, hospitalization; rehosp, rehospitalization.
Note: Dark blue lines highlight admitted days by future rehospitalization while dark blue numbers signify the total such days.

From index CDI admission average length of stay was 16.4 days per annum

Figure 5. Provider costs and estimated reimbursement for top 10 MS-DRGs by hospitalization

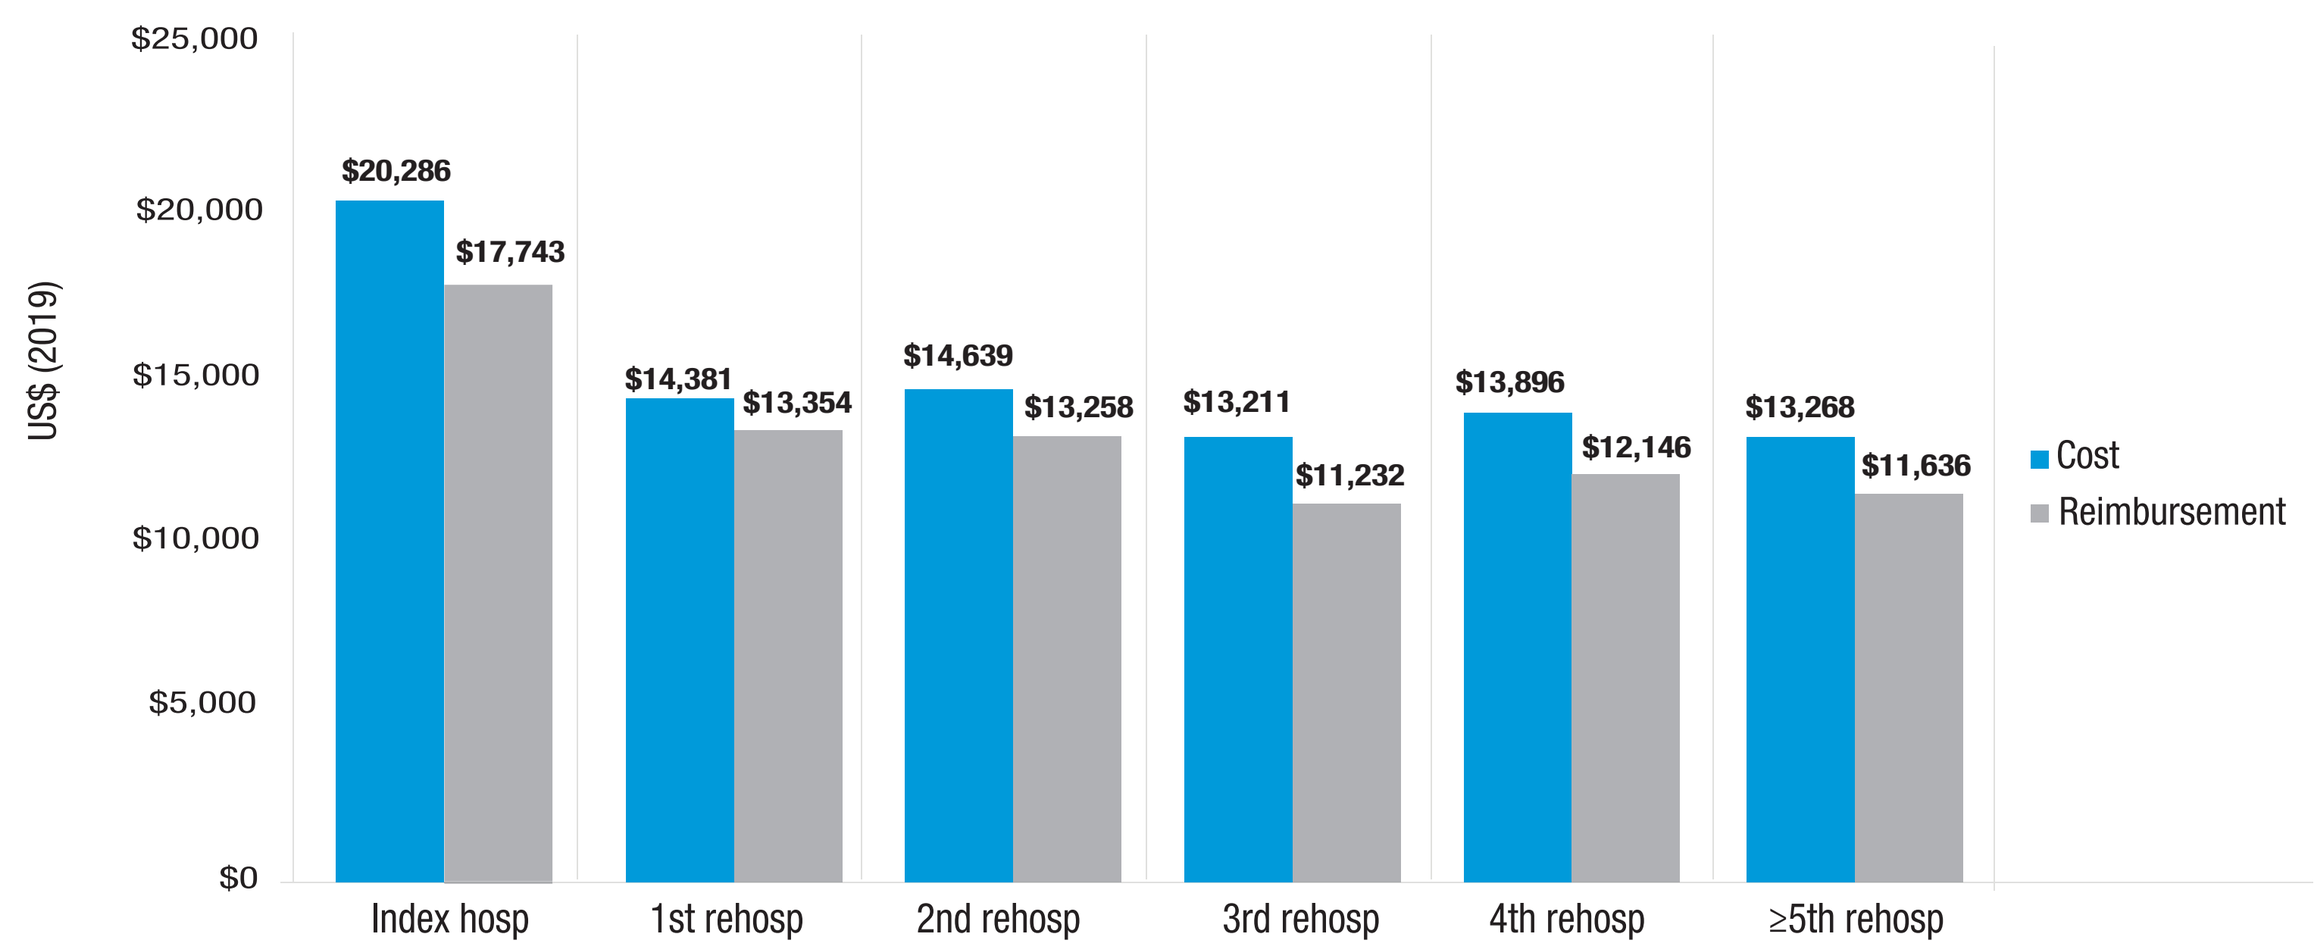
US$ (2019)
$20,286
$17,743
$14,381
$13,354
$14,639
$13,258
$13,211
$11,232
$13,896
$12,146
$13,268
$11,636
Index hosp
1st rehosp
2nd rehosp
3rd rehosp
4th rehosp
≥5th rehosp
Cost
Reimbursement

hosp, hospitalization; MS-DRG, Medicare Severity Diagnosis-Related Group; rehosp, rehospitalization.

At every hospitalization, the cost of providing care exceeded expected reimbursement

Figure 6. Provider cost of care for admissions with ICU involvement by admission type

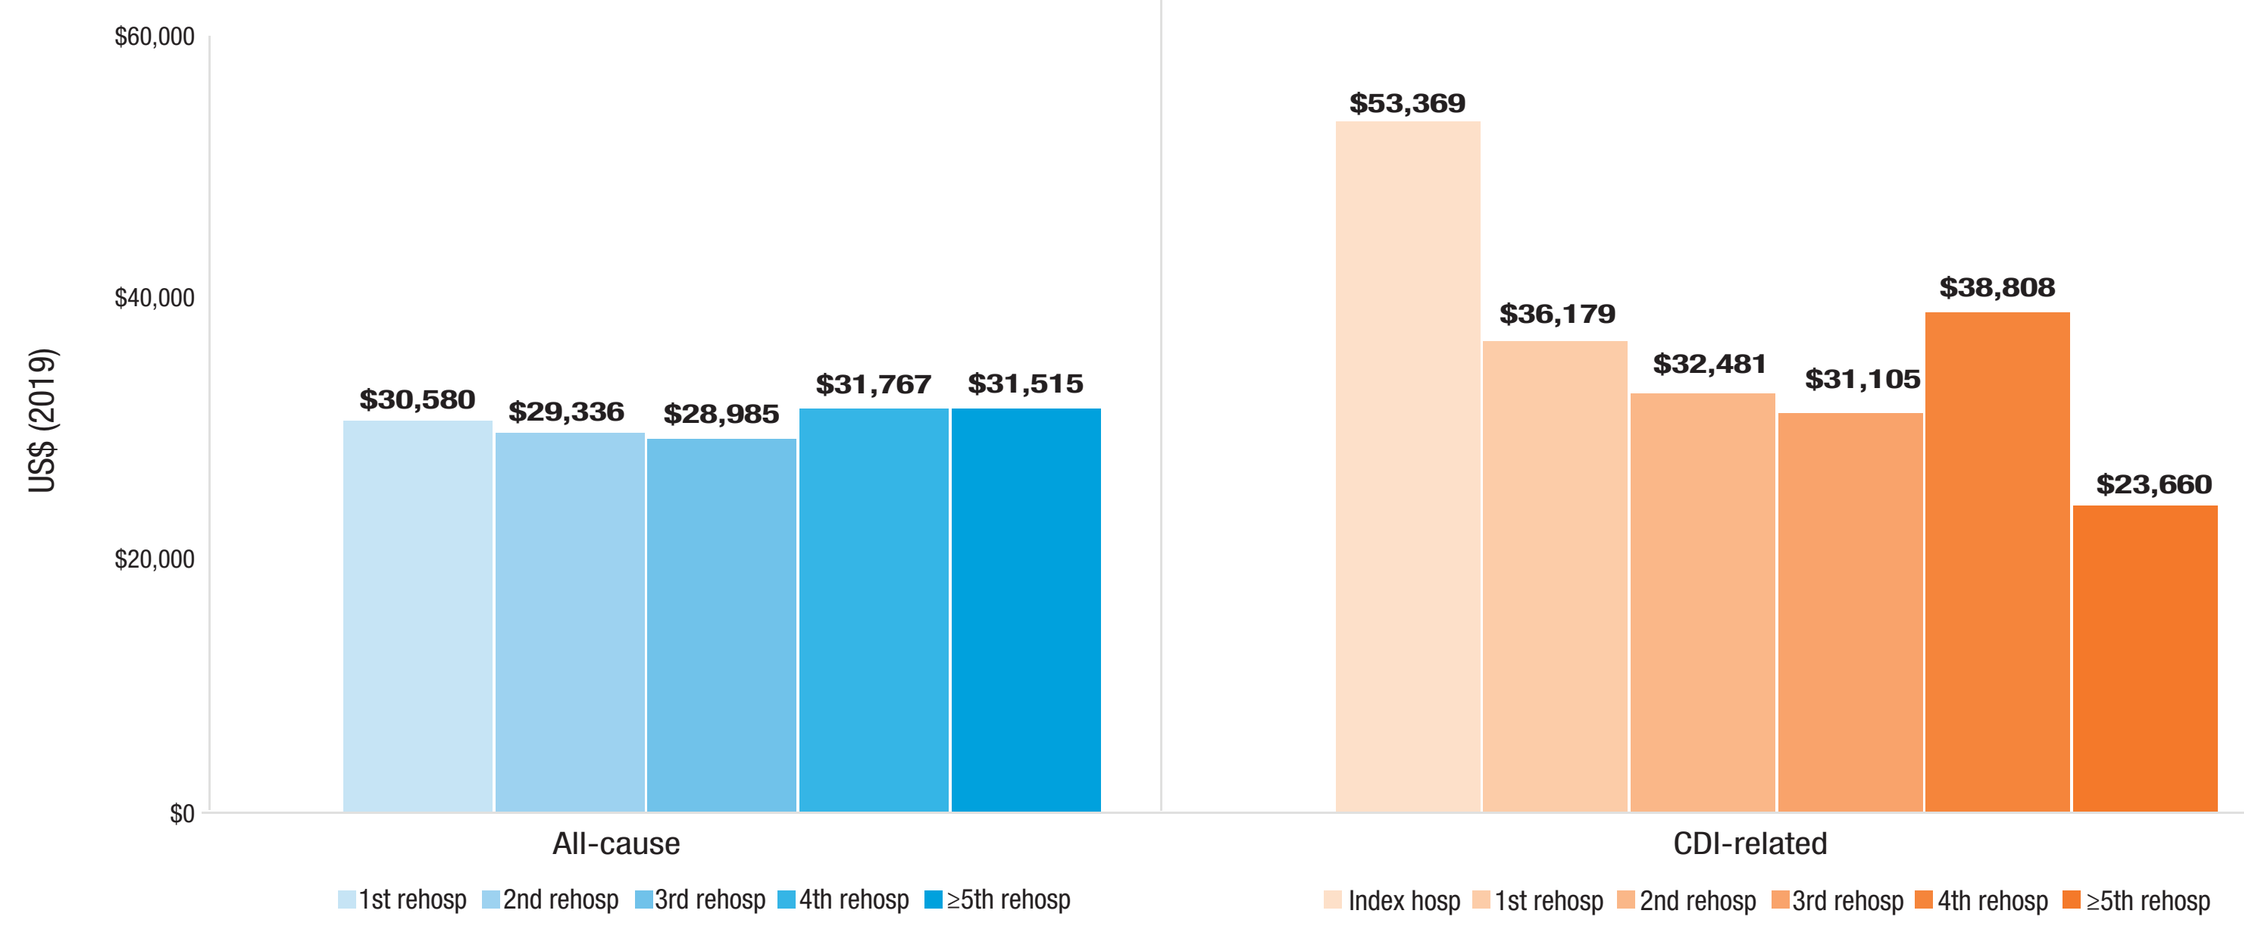
US$ (2019)
$30,580
$29,336
$28,985
$31,767
$31,515
$53,369
$36,179
$32,481
$31,105
$38,808
$23,660
All-cause
CDI-related
1st rehosp
2nd rehosp
3rd rehosp
4th rehosp
≥5th rehosp
Index hosp

CDI, *Clostridioides difficile* infection; hosp, hospitalization; ICU, intensive care unit; rehosp, rehospitalization.

Admissions requiring ICU stays were twice as costly to providers than the average cost

RESULTS

Sample characteristics

- 108,329 patients comprised the final study sample with a CDI-related index hospitalization
- Mean age at index was 66 years (SD 18); mostly female (57.4%); predominatnly white (78.6%); with Medicare the most usual coverage (66.4%)

Primary diagnosis

- MS-DRGs coding for sepsis represented the most common system organ class (**Table 1**)

Rehospitalization

- 42.0% of patients (45,478) were rehospitalized during the 12-month follow-up (**Figure 2**)
- Risk of all-cause readmission increased with each subsequent rehospitalization: 45% following index, 53% following the first rehospitalization, 57% after the second, 58% after the third, and 62% after the fourth readmission
- Beyond the fifth rehospitalization, the mean number of all-cause rehospitalizations was 1.61, while for CDI-related rehospitalizations the mean was 0.71

Mortality

- During the index CDI hospitalization, 6.4% died, and 6.8%, 5.7%, and 4.2% died for first, second, and third or more CDI-related rehospitalization (**Figure 3**)

Length of stay

- The index CDI hospitalization was the longest with 10.3 days on average (**Figure 4**)
- All-cause first through third rehospitalizations were 6.8 days long on average, and fourth or later rehospitalizations were 6.9 days long on average

ICU utilization

- Of the index CDI hospitalizations, 27.6% involved an ICU stay. Of the subsequent all-cause readmissions, 18.4%, 18.5%, 19.3%, 19.5%, and 19.9% required an ICU stay from first through fifth or more, respectively
- The index CDI hospitalizations that involved an ICU stay lasted for 17 days on average, and 7 of which were spent in the ICU
- Subsequent all-cause readmissions with an ICU stay lasted for 10 days, with ICU days averaging 4 days

Complications

- During the index CDI hospitalization, 22.5% of patients had sepsis and 1.6% required bowel surgery
- In the subsequent all-cause rehospitalizations, the sepsis rate varied between 13% and 22%, while the rate of bowel surgeries varied between 2.0% and 0.7%

Economic outcomes

- Based on all MS-DRGs, the average cost for the index CDI hospitalization was $25,691, and cumulative CDI-related hospital costs for patients with 1, 2, 3, and 4+ CDI-related rehospitalizations were $43,601, $60,996, $78,356, and $110,618, respectively

Limitations

- Rehospitalizations are very likely to be underestimated owing to non-index readmissions—a significant continuity of care barrier in US healthcare, constituting 25% of all readmissions[6]
- Inpatient mortality likely substantially underestimates total mortality in this population owing to unobserved mortality in other settings
- Costs related to CDI are likely underestimated because the data only include inpatient hospital admissions, and data from other care settings are not captured
- Clinical depth including severity of disease and other indicators of patient condition are not available

References

1. US CDC. 2018 Annual Report for the Emerging Infections Program for *Clostridioides difficile* infection. Accessed April 4, 2022. https://www.cdc.gov/hai/eip/Annual-CDI-Report-2018.html
2. Feuerstadt P, et al. *SAGE Open Med.* 2021;9:1-8.
3. Lessa FC, et al. *N Engl J Med.* 2015;372:825-834.
4. Feuerstadt P, et al. *J Med Econ.* 2020;23(6):603-609.
5. Premier Applied Sciences®. Premier Inc. Premier Healthcare Database White Paper: Data that informs and performs. https://learn.premierinc.com/white-papers/premier-healthcaredatabase-whitepaper
6. Chappidi MR, et al. *J Urology.* 2017;197(1):235-240.

Acknowledgments

The authors would like to acknowledge Orsolya Lunacsek of Xcenda for assistance with the poster content.

This study was supported by Ferring Pharmaceuticals.
Presented at ISPOR 2022, May 15-18, 2022, Washington, DC.