Abstract #115307
Poster #EE350

Economic Burden of Recurrence Among Resected Early Stage NSCLC Medicare Patients

Jay M. Lee,[1] Rongrong Wang,[2] Ann Johnson,[2] Sarika Ogale,[2] Matthew Kent,[3] Janet S. Lee[2]

[1]David Geffen School of Medicine at UCLA, Los Angeles, CA, USA; [2]Genentech Inc, South San Francisco, CA, USA; [3]Genesis Research, Hoboken, NJ, USA

BACKGROUND

- Early-stage non-small cell lung cancer (eNSCLC) comprises more than half of all cases of NSCLC at diagnosis[1]
- Surgical resection is the primary treatment approach for operable patients with resectable eNSCLC, and adjuvant chemotherapy has been the historical standard of care for metastatic lymph node disease[2]
- However, adjuvant chemotherapy has shown a modest overall survival (OS) benefit in clinical trials,[3] and rates of recurrence are high[4,5]
- Survival outcomes have been shown to worsen with recurrence and advanced disease,[6] but the real-world economic burden of eNSCLC in resected patients with recurrence has not been well characterized
- Newer adjuvant treatment options, including perioperative immune checkpoint inhibitors or targeted therapies, have demonstrated improved disease-free survival, which may reduce the economic burden of resected eNSCLC[7-9]
- With new treatment options for patients with eNSCLC recently approved and recommended,[3] understanding the economic burden of recurrence can help to inform healthcare decision-making in this population
- The objective of this study was to evaluate the real-world incremental healthcare costs associated with recurrence in patients with resected eNSCLC

METHODS

Study Design

- This retrospective observational cohort study used Surveillance, Epidemiology, and End Results cancer registry data linked with Medicare claims data
- Patients aged ≥65 years who were newly diagnosed with NSCLC stages IA to IIIB (American Joint Committee on Cancer [AJCC] Cancer Staging Manual, 7th edition) who underwent surgery between January 2010 and December 2015 were eligible
 - Continuously enrolled in Medicare Parts A, B and D for ≥7 months prior to and ≥12 months after diagnosis (Figure 1)
 - At least 6 months of continuous enrollment were required for surgery-only and neoadjuvant-only groups after the surgery date, and after first adjuvant line of therapy (LOT) for the adjuvant-only and neoadjuvant + adjuvant groups; continuous enrollment in Medicare Parts A, B and D for ≥1 month after the (pseudo-) recurrence date (defined below) was also required
 - Patients with small cell lung cancer, neuroendocrine/carcinoid tumors, large cell carcinoma, missing diagnosis/staging information or enrollment in a health maintenance organization, Veterans Affairs or military hospital were excluded

Figure 1. Study design

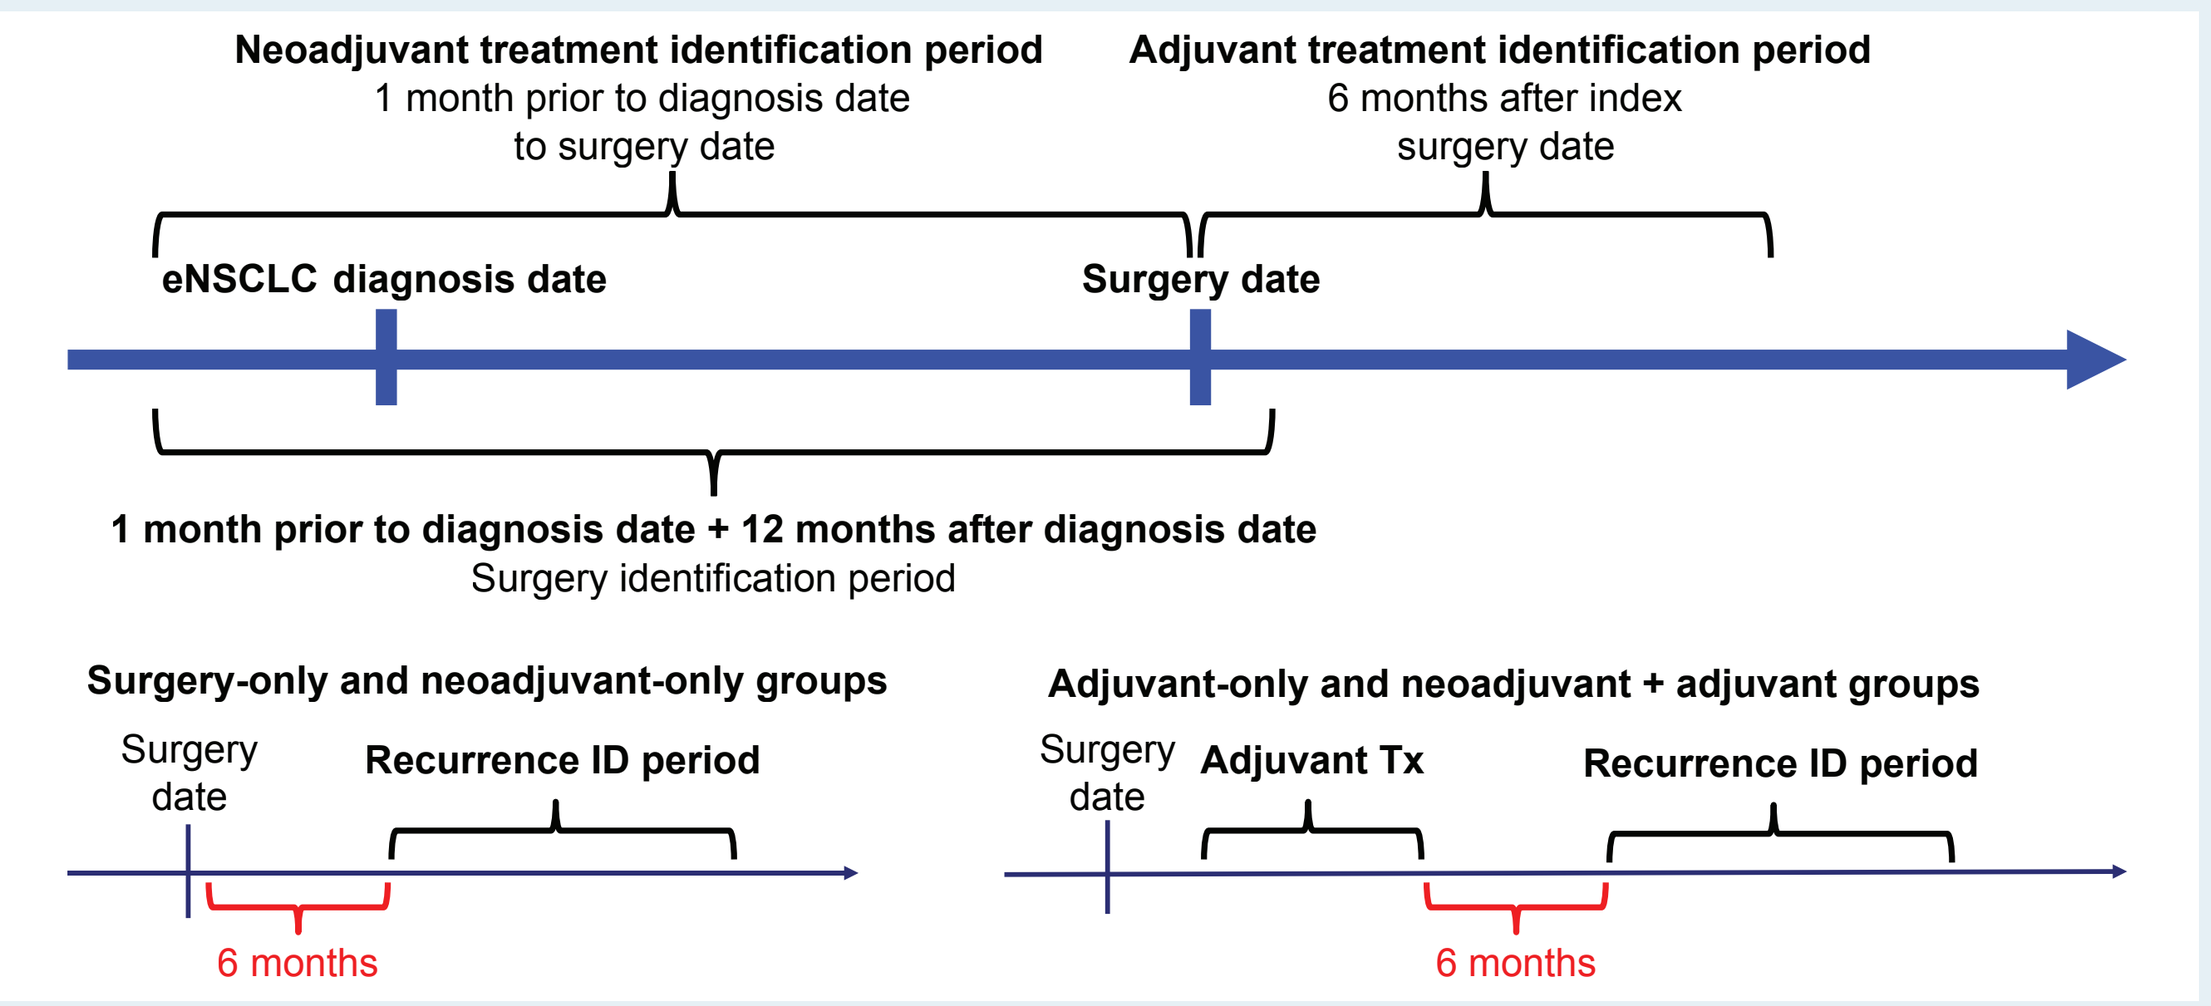
Neoadjuvant treatment identification period
1 month prior to diagnosis date to surgery date
Adjuvant treatment identification period
6 months after index surgery date
eNSCLC diagnosis date
Surgery date
1 month prior to diagnosis date + 12 months after diagnosis date
Surgery identification period
Surgery-only and neoadjuvant-only groups
Surgery date
Recurrence ID period
6 months
Adjuvant-only and neoadjuvant + adjuvant groups
Surgery date
Adjuvant Tx
Recurrence ID period
6 months

Study Cohorts

- Among identified eNSCLC resected patients, we identified two study cohorts:
 - Patients with recurrence
 - Patients without recurrence
- Definition of recurrence
 - Adapted from Hassett 2014[10] and based on clinical expert input; recurrence rates were benchmarked against literature estimates to confirm the robustness
 - Recurrence was defined as evidence of secondary malignant neoplasm (excluding lung), surgery, radiation, chemotherapy, immunotherapy or targeted therapy from 6 months after surgery or end of adjuvant treatment until the end of follow-up (end of enrollment or study period, or death). Treatment definitions are provided in Supplementary Table S1

Study Outcomes

- The study outcomes below were measured starting from 2 months prior to the recurrence date until the end of follow-up. Pseudo-recurrence dates were assigned to non-recurrence patients based on the interval between surgery or adjuvant treatment end date and recurrence date for the matched recurrence patient (see matching algorithm in 'Statistical Analysis' section)
- Direct all-cause costs per patient per month (PPPM)
 - Costs include Medicare-reimbursed amounts and out-of-pocket payments and were adjusted for inflation to 2021 dollars using the medical care component of the Consumer Price Index
- Healthcare resource utilization (HCRU) PPPM
 - Number of inpatient visits, outpatient visits, physician office visits and emergency department (ED) visits

Statistical Analysis

- Descriptive statistics summarized patient characteristics
- Patients with recurrence were matched 1:1 with patients with no recurrence using exact matching on cancer stage and treatment (surgery only, adjuvant only, neoadjuvant only, adjuvant + neoadjuvant), and propensity score matching (PSM) on other characteristics (nearest neighbor approach based on age, sex, race/ethnicity, Charlson Comorbidity Index, histology, lymph node positivity, tumor grade, neoadjuvant/adjuvant treatment)
 - Welch Two Sample t-tests were used to assess differences in PPPM between patients in the matched recurrence vs no-recurrence groups overall for stages IA to IIIB NSCLC
- A sub-analysis comparing costs between recurrence and no-recurrence groups was conducted in patients with stages II to IIIA NSCLC
- Sensitivity analysis compared pre- vs post-recurrence costs in the recurrence group, where each patient served as their own control

RESULTS

Patients

- 6330 eligible patients with surgical resection were included; 2172 (34%) had evidence of recurrence (Figure 2)
- Recurrence rates based on Definition 2 were closest to benchmark rates found in the literature and used in this study. The adapted definition of recurrence for this real-world study population provided a robust patient cohort (Table 1)
- Before matching, patient demographics were largely similar between study cohorts, but recurrence patients were more likely to have higher tumor stage and grade and positive lymph nodes, and to receive adjuvant treatment (Table 2)
- After PSM, 1612 patients with and without recurrence were analyzed (Figure 2)
- All post-matching characteristics were similar between groups (Table 2)
 - Mean age was 74 years, 55%-56% were female, 85%-86% were White and 19% had stage III disease

Figure 2. Patient attrition

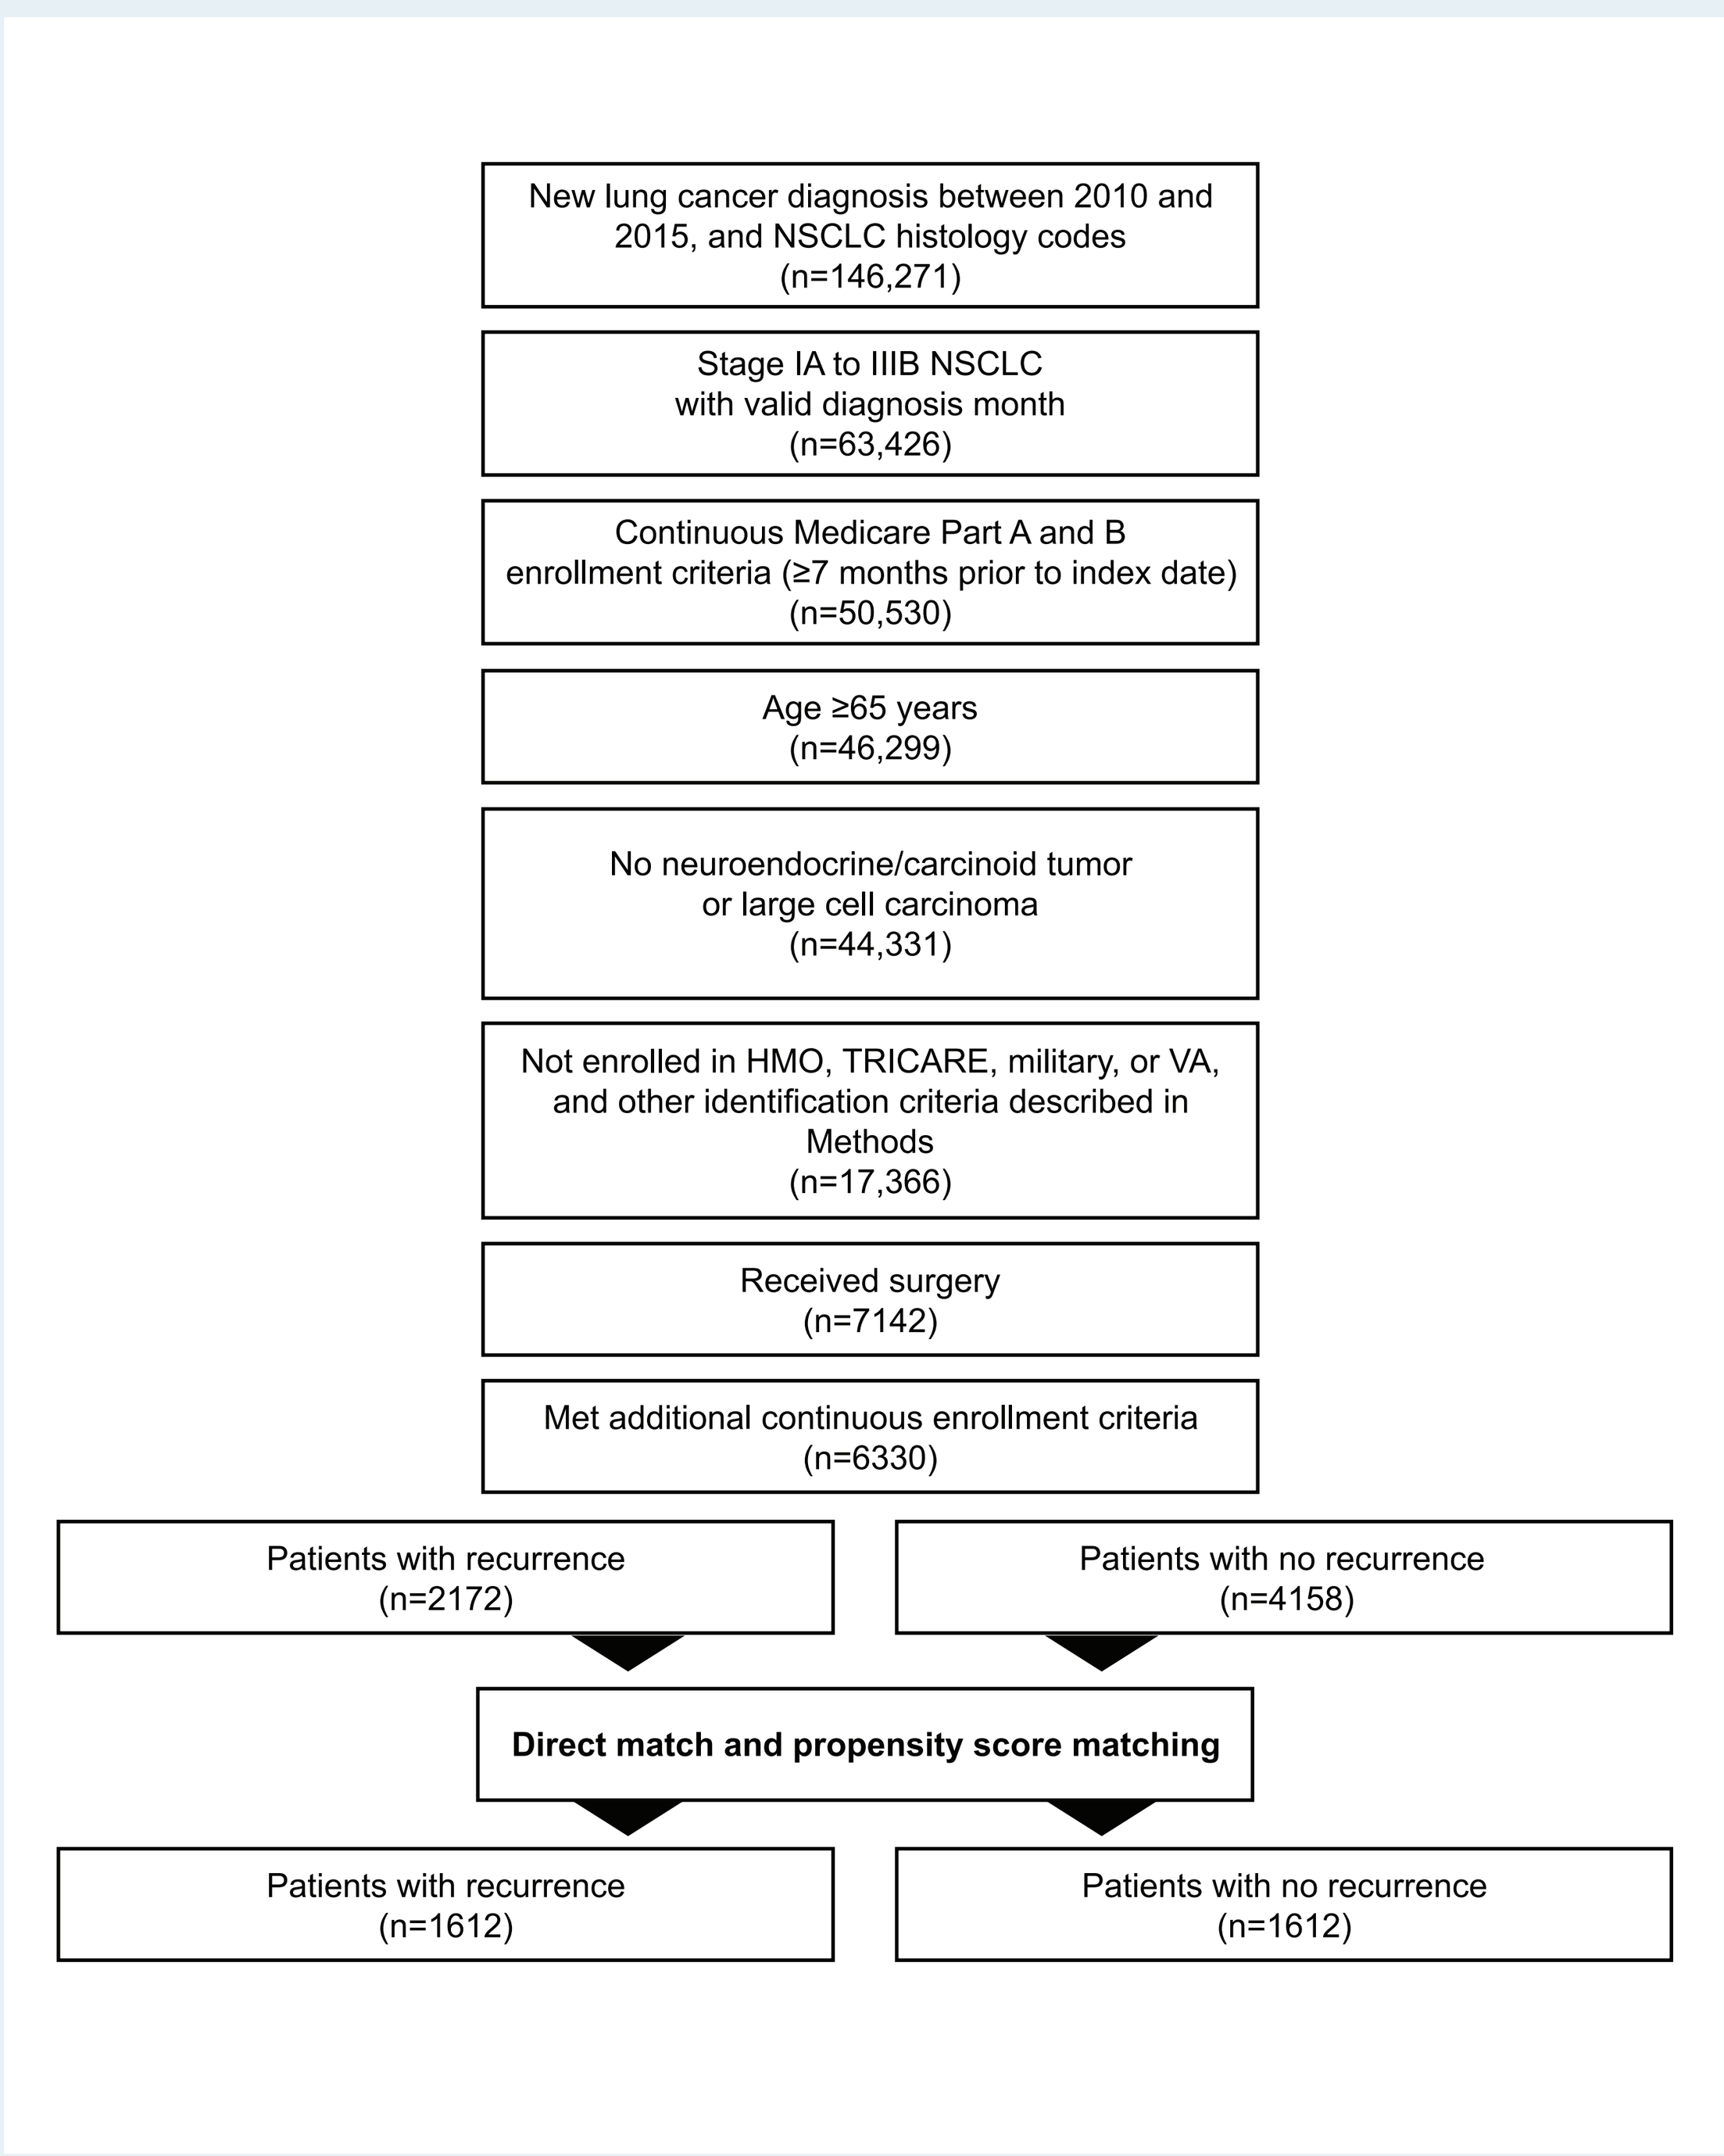
New lung cancer diagnosis between 2010 and 2015, and NSCLC histology codes
(n=146,271)
Stage IA to IIIB NSCLC with valid diagnosis month
(n=63,426)
Continuous Medicare Part A and B enrollment criteria (≥7 months prior to index date)
(n=50,530)
Age ≥65 years
(n=46,299)
No neuroendocrine/carcinoid tumor or large cell carcinoma
(n=44,331)
Not enrolled in HMO, TRICARE, military, or VA, and other identification criteria described in Methods
(n=17,366)
Received surgery
(n=7142)
Met additional continuous enrollment criteria
(n=6330)
Patients with recurrence
(n=2172)
Patients with no recurrence
(n=4158)
Direct match and propensity score matching
Patients with recurrence
(n=1612)
Patients with no recurrence
(n=1612)

VA, Veterans Affairs.

Table 1. Incremental adaptation of recurrence rate definition

	Definition 1	Definition 2	Definition 3
Criteria	• At least 1 inpatient claim or 2 outpatient claims with diagnosis codes for secondary malignant neoplasm (excluding lung)	• At least 1 inpatient claim or 2 outpatient claims with diagnosis codes for secondary malignant neoplasm (excluding lung) • Surgery based on procedure codes • Non-surgical treatment (radiation, chemo, IO, TT) based on procedure/drug codes	• At least 1 inpatient claim or 2 outpatient claims with diagnosis codes for secondary malignant neoplasm (excluding lung) • Surgery based on procedure codes • Non-surgical treatment (radiation, chemo, IO, TT) based on procedure/drug codes • Diagnosis code for secondary malignant neoplasm of lung • Palliative care based on diagnosis codes • Biopsies based on procedure codes
5-year Recurrence Rate	**19%**	**41%**	**78%**
Recurrence events by stage, n (%)	996 (14)	2248 (31)	4286 (60)
IA	211 (8)	639 (23)	1720 (62)
IB	282 (13)	642 (30)	1288 (61)
II	180 (22)	309 (38)	587 (60)
IIIA	192 (28)	294 (42)	412 (59)
IIIB	131 (17)	264 (35)	379 (50)

Hassett 2014: Validation chart abstraction study of secondary malignant neoplasm and chemotherapy codes as indicators of recurrence after definitive local therapy for stage I-III lung cancer using Cancer Care Outcomes Research and Surveillance Consortium (CanCORS)/Medicare and Health Maintenance Organization (HMO)/Cancer Research Network. To avoid chemotherapy given for adjuvant treatment, chemotherapy identified within 6 months of diagnosis was excluded.
Red text indicates changes between definitions.

Table 2. Pre- and post-matching patient characteristics

Characteristic	Pre-matching groups			Post-matching groups		
	Recurrence (n=2172)	No recurrence (n=4158)	SMD	Recurrence (n=1612)	No recurrence (n=1612)	SMD
Age at diagnosis, mean (SD), years	73.7 (5.6)	73.4 (5.5)	0.05	73.7 (5.7)	73.8 (5.6)	0.02
Female, %	56%	59%	0.07	56%	55%	0.02
Race/ethnicity, %			0.10			0.05
White	85%	86%		86%	85%	
Black	6%	6%		6%	6%	
Asian	5%	4%		5%	5%	
Other	2%	2%		2%	2%	
Hispanic	1%	1%		1%	1%	
Native American	0.2%	0.2%		0.3%	0.2%	
Missing	0.1%	1%		0.1%	0.1%	
Median income, %[a]			0.07			0.06
≤$40,502	21%	21%		20%	21%	
$40,503 to $56,178	22%	22%		22%	21%	
$56,179 to $79,560	26%	24%		25%	27%	
>$79,560	25%	27%		27%	25%	
Missing	7%	6%		7%	7%	
College education, %[a]			0.04			0.05
≤14%	20%	20%		20%	20%	
15% to 25%	25%	24%		24%	25%	
26% to 42%	23%	24%		24%	23%	
>42%	25%	26%		26%	25%	
Missing	7%	6%		7%	7%	
Charlson Comorbidity Index score at baseline, mean (SD)	2.3 (1.9)	2.1 (1.8)	0.09	2.2 (1.9)	2.3 (2.0)	0.05
Tumor stage (AJCC 7th ed), %			0.48			<0.001
IA	29%	48%		31%	31%	
IB	23%	25%		24%	24%	
II	24%	16%		26%	26%	
III[b]	–	–		19%	19%	
IIIA	22%	10%		–	–	
IIIB	2%	1%		–	–	
Tumor grade, %			0.28			0.05
1	11%	19%		10%	10%	
2	44%	45%		45%	45%	
3	36%	27%		35%	37%	
4	1%	1%		1%	1%	
Missing	8%	8%		8%	8%	
Histology, %			0.07			0.07
Adenocarcinoma, NOS	44%	41%		42%	45%	
Squamous cell carcinoma, NOS	29%	29%		30%	29%	
Other	27%	30%		28%	26%	
Positive Lymph Node Resection, %			0.40			0.08
No	61%	78%		65%	63%	
Yes	27%	13%		25%	24%	
No nodes examined	12%	9%		10%	13%	
Unknown	0.3%	0.3%		1%	0.4%	
Treatment cohort, %			0.50			<0.001
Surgery only	52%	74%		53%	53%	
Neoadjuvant only	4%	4%		4%	4%	
Adjuvant only	38%	19%		37%	37%	
Neoadjuvant + adjuvant	7%	3%		6%	6%	

SMD <0.1 was used to determine if covariate was considered balanced between groups.
NOS, not otherwise specified; SD, standard deviation; SMD, standardized mean difference.
[a] Based on US Census tract aggregate data, not patient-level data.
[b] Categorized as stage III for post-matching patients.

Costs of Recurrence

- Over an average follow-up period of 30 months, total all-cause healthcare costs were nearly 5-fold higher (+$4577 PPPM; *P*<0.001) for patients with stage IA to IIIB NSCLC with recurrence after matching for observable characteristics between groups (Figure 3)
- All component costs were significantly higher in the recurrence group, with the greatest difference in inpatient costs (+$2181 PPPM; Figure 3)

Figure 3. Comparison of PPPM costs for recurrence vs no recurrence in patients with stage IA-IIIB NSCLC

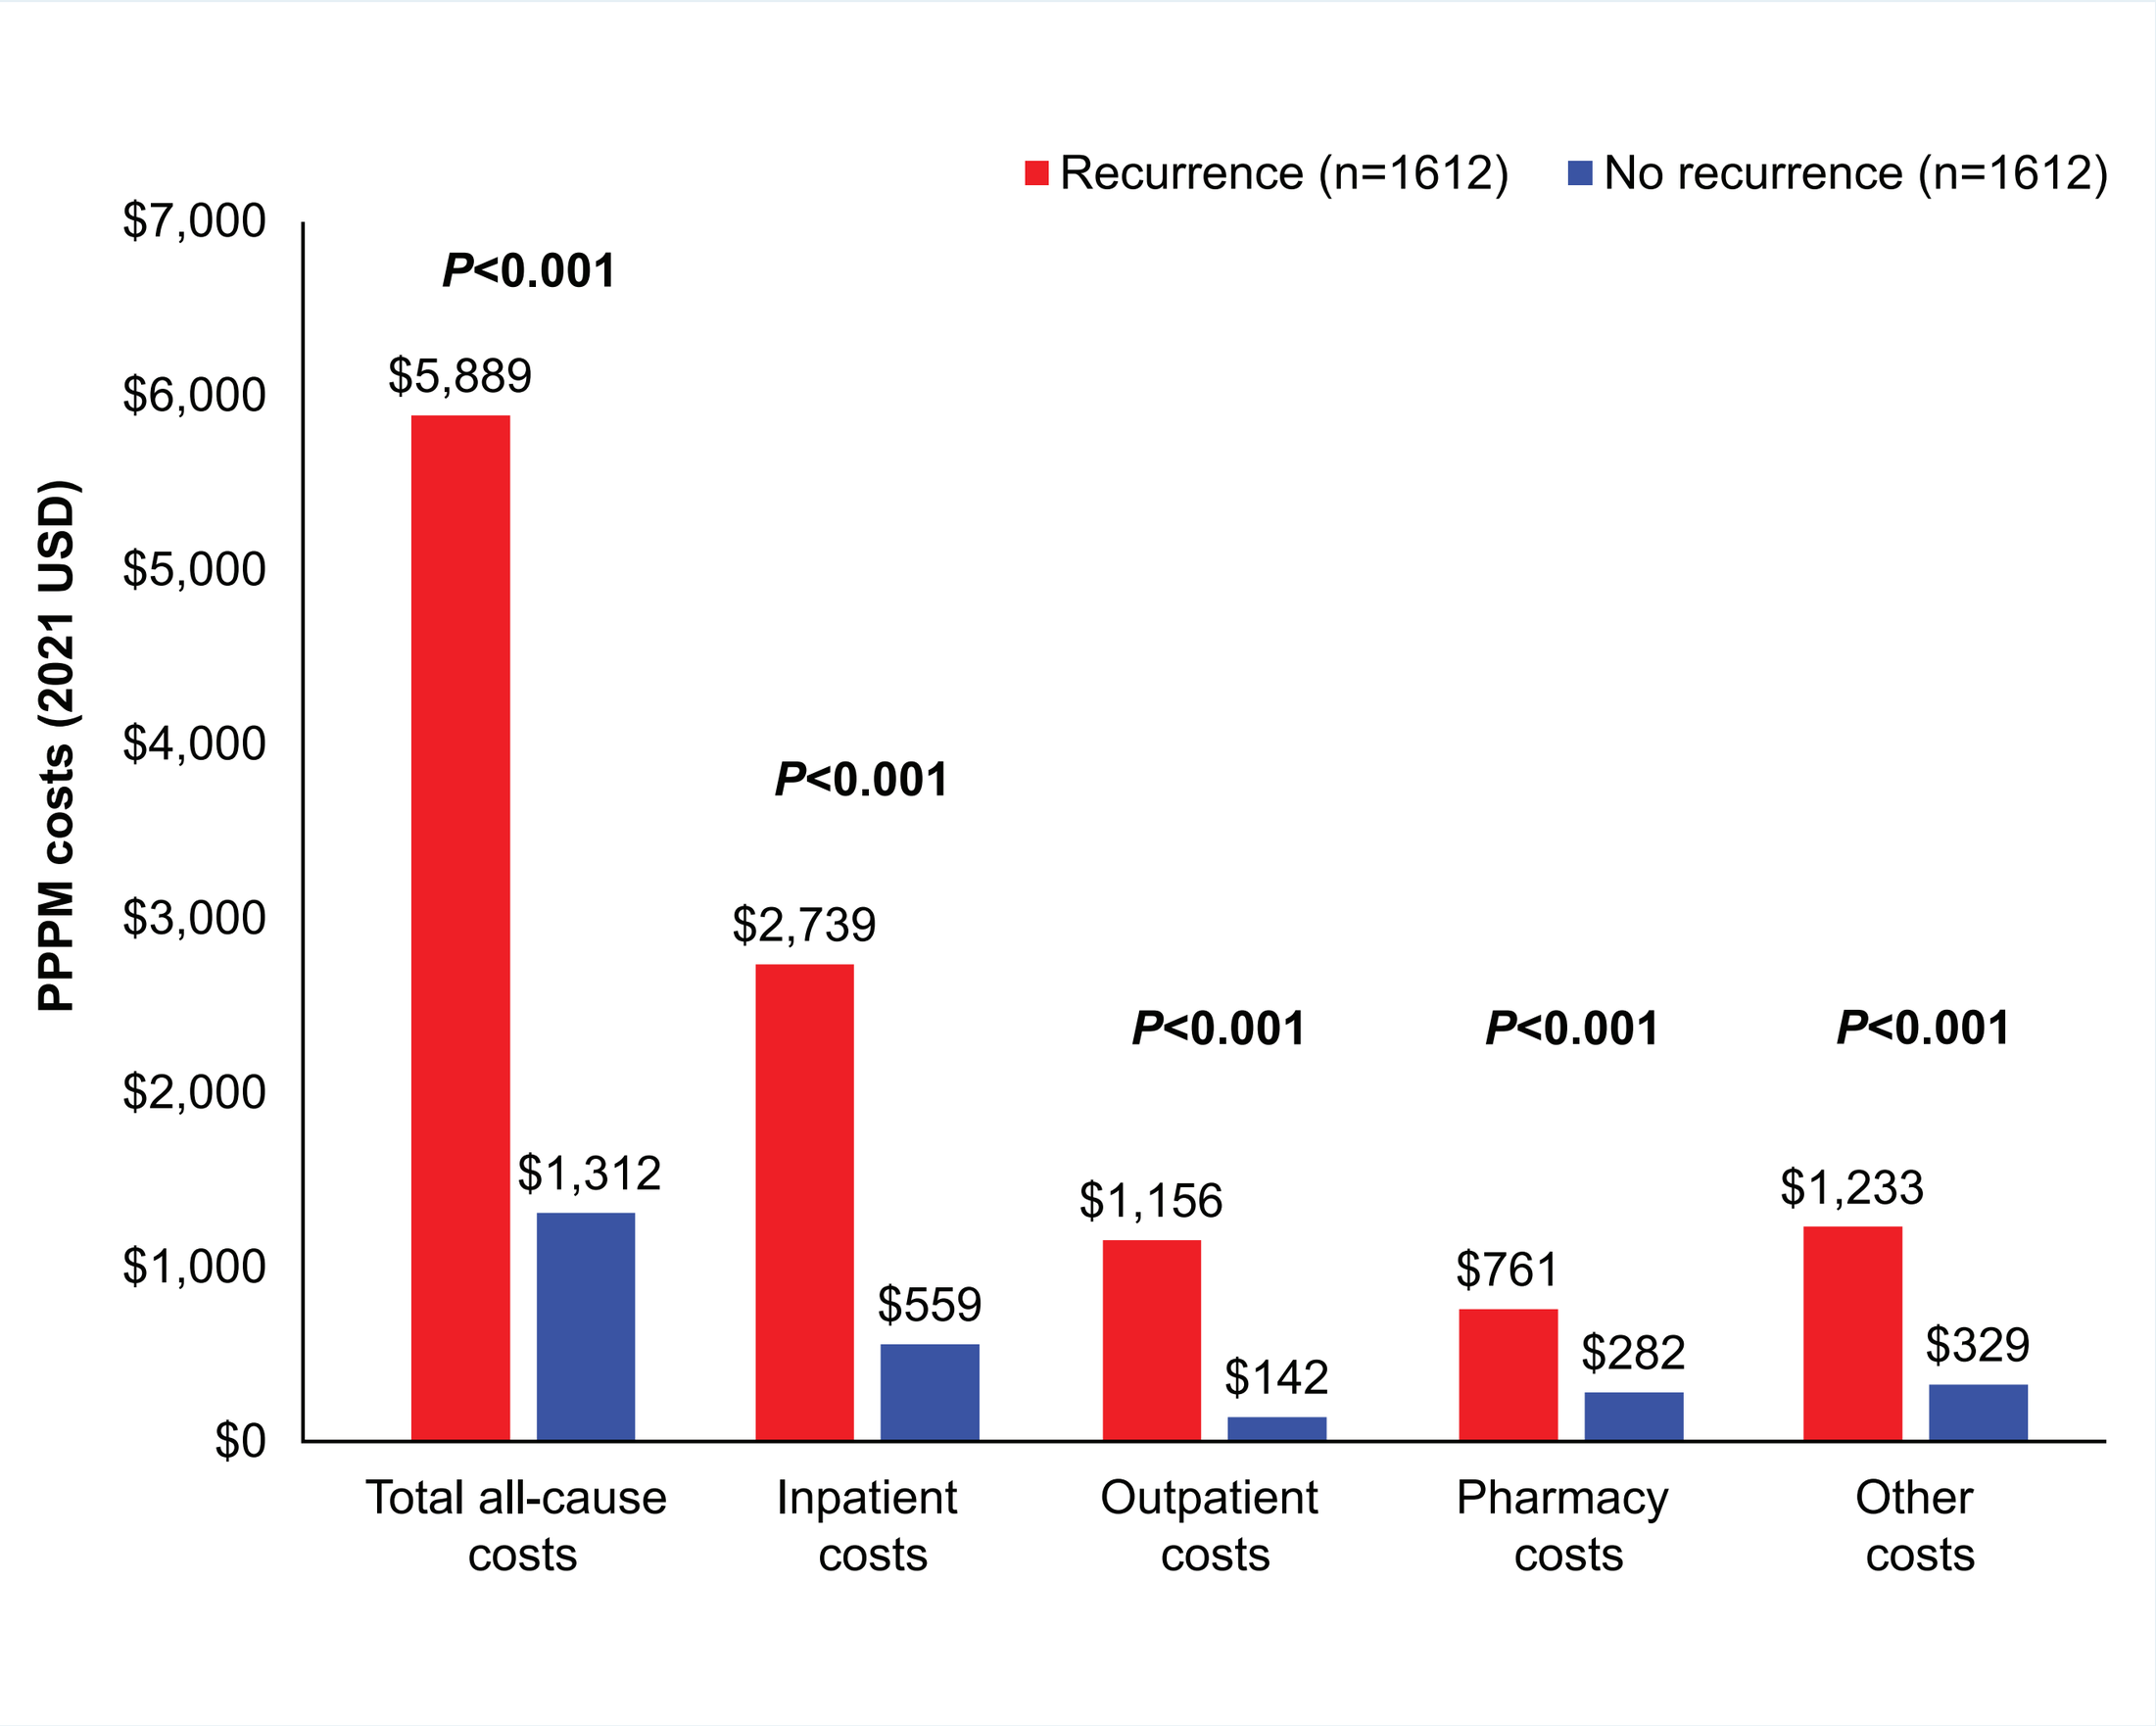
Recurrence (n=1612)
No recurrence (n=1612)
PPPM costs (2021 USD)
$7,000
$6,000
$5,000
$4,000
$3,000
$2,000
$1,000
$0
P<0.001
$5,889
$1,312
Total all-cause costs
P<0.001
$2,739
$559
Inpatient costs
P<0.001
$1,156
$142
Outpatient costs
P<0.001
$761
$282
Pharmacy costs
P<0.001
$1,233
$329
Other costs

- Total healthcare costs were higher for patients with recurrence among those with stage II-IIIA eNSCLC (*P*<0.001; Figure 4)
- Analysis of pre- vs post-recurrence costs for patients with recurrence were consistent with the primary findings, with a difference in total all-cause costs of +$5927 PPPM post recurrence (*P*<0.001)

Figure 4. Comparison of PPPM costs for recurrence vs no recurrence in matched patients with stage II-IIIA NSCLC

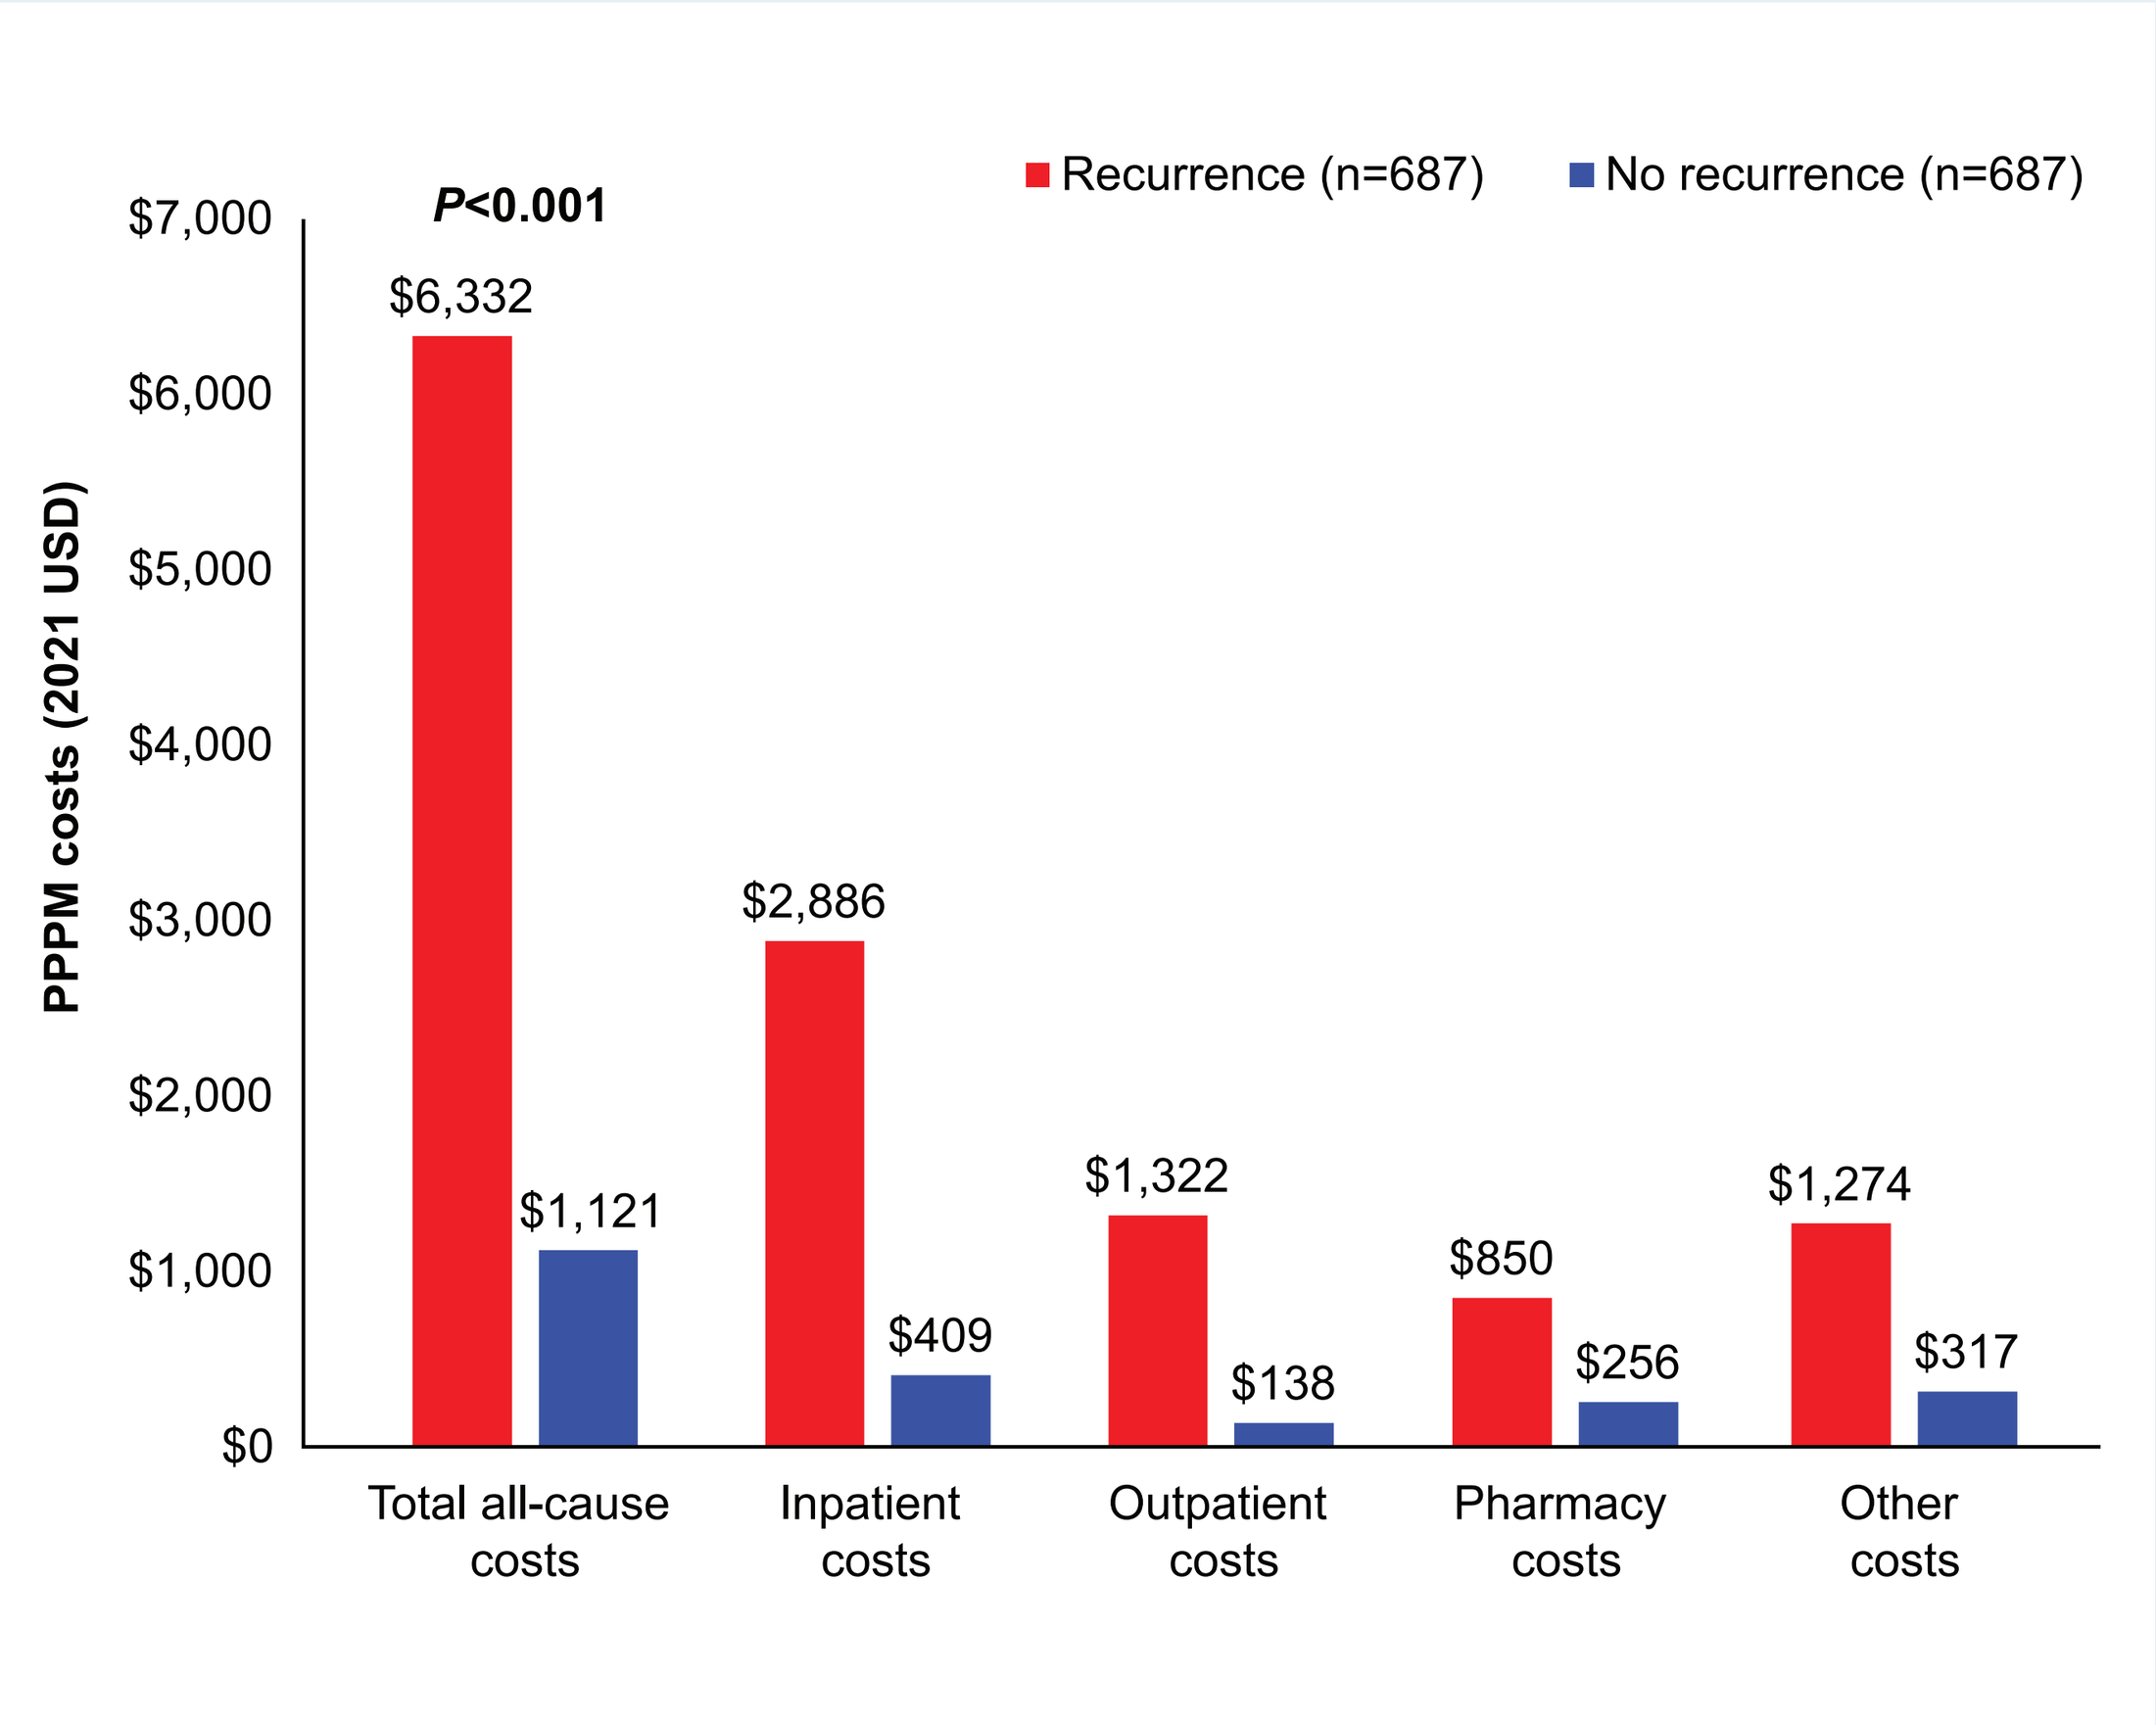
Recurrence (n=687)
No recurrence (n=687)
PPPM costs (2021 USD)
$7,000
$6,000
$5,000
$4,000
$3,000
$2,000
$1,000
$0
P<0.001
$6,332
$1,121
Total all-cause costs
$2,886
$409
Inpatient costs
$1,322
$138
Outpatient costs
$850
$256
Pharmacy costs
$1,274
$317
Other costs

Healthcare Resource Utilization

- Patients with recurrence had significantly greater HCRU across all measures, including mean inpatient and outpatient visits, physician office visits and ED visits PPPM, than matched patients with no recurrence (Table 3)

Table 3. Comparison of PPPM HCRU for recurrence vs no recurrence in matched patients with stage IA-IIIB NSCLC

HCRU, mean (SD), PPPM	Recurrence (n=1612)	No recurrence (n=1612)	Difference	*P* value
Inpatient visits	0.19 (0.24)	0.08 (0.12)	0.11	*P*<0.001
Outpatient visits	0.98 (0.93)	0.33 (0.37)	0.65	*P*<0.001
Physician office visits	3.66 (2.64)	1.35 (1.32)	2.30	*P*<0.001
ED visits	0.20 (0.25)	0.09 (0.11)	0.12	*P*<0.001

CONCLUSIONS

- Based on a real-world population, recurrence in patients with resected eNSCLC is associated with significantly higher all-cause healthcare utilization and costs
 - Over an average 30 months of follow-up from (pseudo-) recurrence date, total healthcare costs were approximately 5 times higher for eNSCLC patients with recurrence vs patients without recurrence
- Sensitivity analysis of pre- vs post-recurrence costs was consistent with the primary findings
- Some limitations should be noted:
 - Study findings may not be generalizable to a non-Medicare population
 - Recurrence was indirectly identified based on claims data and not formally validated, which may have led to potential misclassification because many elderly patients decline or are not offered treatment after recurrence
 - Propensity score matching does not control for unobservable differences between patient cohorts, which could potentially bias results due to confounding
- New therapeutic options with adjuvant immune checkpoint inhibitors or targeted therapies[7-9] that decrease recurrence may reduce the economic burden associated with eNSCLC

REFERENCES

1. Lu T, et al. *Cancer Manag Res.* 2019;11:943-53.
2. Pisters K, et al. *J Clin Oncol*. Published online February 15, 2022.
3. Pignon J-P, et al. *J Clin Oncol.* 2008;26:3552-9.
4. NSCLC Meta-Analysis Collaborative Group. *Lancet.* 2014;383:1561-71.
5. Uramoto H, Tanaka F. *Transl Lung Cancer Res.* 2014;3:242-9.
6. Goldstraw P, et al. *J Thorac Oncol.* 2016;11:39-51.
7. Felip E, et al. *Lancet.* 2021;398(10308):1344-57.
8. Wu Y-L, et al. *N Engl J Med.* 2020;383(18):1711-23.
9. Merck & Co., Inc. Accessed March 15, 2022. https://www.merck.com/news/mercks-keytruda-pembrolizumab-showed-statistically-significant-improvement-in-disease-free-survival-versus-placebo-as-adjuvant-treatment-for-patients-with-stage-ib-iiia-non-small-cell/
10. Hassett MJ, et al. *Med Care.* 2014;52(10):e65-73.

DISCLOSURES

- JML received research support for this study provided by Genentech Inc, a member of the Roche Group
- RW, AJ, SO and JSL are employees and stockholders of Genentech Inc, a member of the Roche Group
- MK is an employee of Genesis Research, which received research funding for this study from Genentech Inc, a member of the Roche Group

ACKNOWLEDGMENTS

- This study is sponsored by Genentech Inc, a member of the Roche Group
- Support for third-party writing assistance for this manuscript, furnished by Jeff Frimpter, MPH, of Health Interactions, was provided by Genentech Inc, a member of the Roche Group

For questions or comments on this poster, please contact Janet Lee at lee.janet@gene.com
Copies of this e-poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without written permission of the authors

International Society for Pharmacoeconomics and Outcomes Research (ISPOR) Annual Meeting 2022 – May 15-18, 2022 – Washington, DC, USA and Virtual