Adherence Advantages of Long-Acting Injectable Antipsychotics (LAIs) in Schizophrenia. Is the Currently Available Real-World Evidence Adequate or Conclusive?

Amna M. Aslam, MBA, Rajesh Nayak,PhD
College of Pharmacy and Health Sciences, St. John's University, Queens, NY 11439.

BACKGROUND

Long-acting injectable (LAI) antipsychotics (APs) have been around a long time. However, their popularity over the past decade has grown significantly because of the many therapeutic opportunities they present in the management of Schizophrenia and other psychotic disorders. Despite their widespread use, LAIs appear to lack a strong evidence-base to conclusively demonstrate their Real-World Evidence (RWE) advantages of adherence, particularly with respect to the newer generation of LAIs.

OBJECTIVES

The current study aims to conduct a systematic review of relevant literature to gather RWE to document LAI's effectiveness in achieving improved medication adherence based on the data obtained from traditional reviews, RCTs, experimental trials, secondary data analyses and observational research.

METHODS

A comprehensive review of literature (2010-present) was undertaken by two reviewers with articles retrieved from Embase, PubMed/Medline, EBSCO, ScienceDirect, PsycINFO, and Cochrane Library databases, employing appropriate adherence and utilization-specific search terms for LAI use in schizophrenia. Selected articles were classified according to the type of data sourced (primary, secondary,patient-reported, observational, etc.), research design employed to generate RWD (RCT, cohort & case-control, mirror-image studies, and others), and effectiveness outcomes that were targeted (primary adherence outcome and other).

RESULTS

The current study utilized n=37 studies meeting the selection criteria, and were classified based on the type of data (primary vs. secondary) in which studies were also organized by the type of Real World Data (RWD), as represented by Randomized Control Trials (RCT), cohort & case-control, mirror-image studies, retrospective observational studies etc. Different types of LAIs featured in the studies included Aripiprazole (Monohydrate, Lauroxil, 1M), Paliperidone Palmitate (4w,12w, 1M, 3M), Fluphenazine (Decanoate), Haloperidol (Decanoate), Olanzapine (Pamoate), Zuclopenthixol, and Risperidone along with Oral Antipsychotic (OAPs) types in conjunction with LAI formulation variations. First & Second Generation Antipsychotics (FGAs & SGAs) were compared with OAPs in many studies with respect to the targeted outcome of adherence, as well as psychiatric outcomes associated with relapse, rehospitalization, and health care utilization. Adherence methods employed included primarily Proportion of Days Covered (PDC), measures obtained from claims-based prescription refill records, such as time until initial nonadherence, Discontinuation Rates, and Medication Gap. Psychiatric rating tools included scales like Clinical Global Impression (CGI) Scale, Global Assessment of Functioning (GAF), Brief Psychiatric Rating Scale (BPRS), among other things. A decrease in hospitalization and/or rehospitalization rates was noted in n= 22 studies which corresponded to a significant decrease in relapse rates as well (n=13). Significant adherence advantages (n=9) and meaningful adherence improvement rates (n=19) were documented as well, with rates varying between 30.90%-82.0% in the studies reviewed.

Table 1: Real World Evidence Focused on Adherence Outcomes

Type of RWE	Setting type	Number of Studies and Study Participants (Range)	LAIs Featured	Adherence Measures Employed
Claims and billing activities	Inpatient and outpatient	# of Studies: 8 Participant Range: 499-15979	Oral risperidone or oral paliperidone to LAI PP1M,PP injections,LAI antipsychotic generation (first-generation [FGA] or second-generation [SGA] antipsychotics), and individual LAI agent (fluphenazine decanoate, haloperidol decanoate, risperidone LAI, and paliperidone palmitate) vs Oral antipsychotics	Paid medical claims (inpatient and outpatient), and pharmacy claims & prescription refill records
Product and disease registries	Mental health units,outpatient pharmacy ,outpatient visits, inpatient visits, and hospitalizations.	# of Studies: 4 Participant Range:180-676	Risperidone long-acting injection, paliperidone palmitate, fluphenazine decanoate , aripiprazole extended-release injectable , zuclopenthixol decanoate , and olanzapine extended release	Proportion of Days Covered (PDC)
Patient-generated data including in home-use settings	Day hospital, part-time therapy center, sheltered housing, protected employment center,pragmatic, open-label, observational, parallel group study,naturalistic conditions study	# of Studies: 6 Participant Range: 25-13087	Risperidone long-acting injectable (RLAI) LAIs and oral antipsychotic, comparisons between LAIs and oral antipsychotics and between FGA-LAIs and SGA-LAIs, FGA-LAI vs SGA-LAI. Haloperidol, Fluphenazine, Zuclopenthixol, Risperidone, Paliperidone, Olanzapine	Clinical Global Impression (CGI) scale and Global Assessment of Functioning (GAF) BPRS 24-items, GAF, Lunsers, haematic samples estimation, body-mass-index ICD-10 diagnosis of Schizophrenia or like Delusional Disorder

Table 2: : Comparison LAIs vs OAPs in Studies

LAIs Examined	First or Second Generation	OAPs compared to LAIs? YES/NO	OAPs Used for Comparison	Authors	Key Summary Findings
Aripiprazole (Monohydrate, Lauroxil, 1M)	SGA	13 studies = YES 1 study= NO	Corresponding Oral Formulation	**14 Studies:** Taylor, et al, Lin, et al, 2019, Marcus, et al, 2015,Lytle, et al, 2017,McCreath, et al 2021,Fricchione, et al, 2017, ,Garcia, et al,2020, Orietta et al, Xue et al,2019 El Khoury et al,2019, Dominic et al, 2017, Sylvain et al,2016, Dasom, et al,2020, Tajima et al, 2019.	Associated with likelihood of PDC ≥80%, LAIs showed a reduction of treatment and hospitalizations vs OAPs. PP1M and PP3M showed superior outcomes when compared to A1M and biweekly LAIs, Improvement with SGA LAI's is noted especially in discontinuation. Medicaid patients with schizophrenia on SGA LAI's had better adherence than those being treated with OAPs.
Paliperidone Palmitate (4 weeks/12w, 1Month/ 3M)	SGA	20 studies = YES 3 studies = NO	Olazapine Extended Release, Aripiprazole Extended Release,Oral Risperidone, Oral Paliperidone	**23 Studies;**Malik et al, 2017,Steve et al 2012,Lin, et al, 2019 ,Marcus, et al, 2015,Lytle, et al, 2017,McCreath, et al 2021,Fricchione, et al, 2017, ,Garcia et al,2020,Patel, et al 2021,Orietta et al, 2018,Nielsen et al, 2014,Lin et al, 2019,,Xue et al,2019,El Khoury et al,2020, Vincent et al,2017,Pilon et al, 2017, Chan et al,2021, Joshi et al,2016, Druais et al,2016, Lee et al, 2021, El Khoury et al,2019, Emond et al,2018,Tajima et al, 2019.	Associated with likelihood of PDC ≥80%. Patients using SGA showed lower odds of nonadherence, discontinuation, and schizophrenia related hospitalizations. Improvement of clinical outcomes and adherence in the youth with LAI side effects appearing similar to OAPs. Improvement with SGA LAI's is noted especially in discontinuation. Medicaid patient with schizophrenia on SGA LAI's had better adherence than those being treated with OAPs. Pharmacy costs from claims data shows PP1M has superiority when treating schizophrenia patients. Transitioning from Oral Ris/ Pali to PP1M improved schizophrenia related HRU and may provide potential cost savings in VHA patients. With the transition to PP3M, medical costs such as ER visits, and inpatient costs decreased which pharmacy costs and adherence to other anti-psychotics increased.
Fluphenazine (Decanoate)	FGA & SGA	6 studIES = YES 3 studies = NO	Olazapine Extended Release Aripiprazole Extended Releasel	**9 Studies;** Marcus, et al, 2015, Offord et al, 2012, Lytle, et al, 2017, McCreath et al 2021,Fricchione et al, 2017,Patel et al 2021,Orietta et al, 2018,Nielsen et al, 2014, ,Lang et al,2012	Patients using SGA showed decrease of nonadherence, discontinuation and schizophrenia related hospitalizations. Improvement of clinical outcomes and adherence in the youth with LAI side effects appearing similar to OAPs.SGA LAIs cost and patient centered outcomes should be taken in account as SGA LAIs are not superior to FGA's regarding duration of hospitalization, and psychiatric hospitalizations.
Haloperidol (Decanoate)	FGA & SGA	10 studies = YES 3 studies = NO	N/A	**13 Studies;** Malik et al, 2017, Offord et al, 2012, Marcus, et al, 2015, Viala et al,2012, McCreath, et al 2021, Fricchione, et al, 2017, ,Garcia, et al,2020, Orietta et al,, Nielsen et al, 2014, ,Lang et al,2012, Lin, et al, 2019, Chan et al,2021,Druais et al,2016, Lee et al, 2021	Patients using SGA showed little to no nonadherence, discontinuation, and schizophrenia related hospitalizations. It was noted SGA LAIs are not superior to FGA's regarding duration of hospitalization, and psychiatric hospitalizations.
Risperidone	SGA	20 studies = YES 5 studies = NO	N/A	**25 Studies;** Kaplan et al,2013, Maliik et al, 2017, Offord et al, 2012, Lin, et al, 2019, Marcus, et al, 2015, Lytle, et al, 2017,, Viala et al,2012, McCreath, et al 2021,Fricchione, et al, 2017,,Schooler, et al,2012,Salgueiro et al,2020 Patel, et al 2021,Orietta et al, 2018,Nielsen et al, 2014,Lin et al, 2019,Bensouda et al,2012, Xue et al,2019,El Khoury et al,2020, Lang et al,2012 Pilon, et al,2017 Chan et al,2021, Joshi et al,2016,Druais et al,2016, , Lee et al, 2021, El Khoury et al,2019	Patients using SGA showed lower odds of nonadherence, discontinuation and schizophrenia related hospitlizations.120 patients were compared in a cohort where improvement of adherence was noted after receiving Risperidone LAI. Participants taking SGA LAI's gained weight especially with Olanzapine LAI. Decrease in medical costs were noted when switched to PP1M with an increase of pharmacy costs which results in similar total costs. SGA LAIs cost and patient centered outcomes should be taken in account as SGA LAIs are not superior to FGA's regarding duration of hospitalization, and psychiatric hospitalizations. Improvement with SGA LAI's is noted especially in discontinuation. Medicaid patient with schizophrenia on SGA LAI's had better adherence than those being treated with OAPs. Pharmacy costs from claims data shows PP1M has superiority when treating schizophrenia patients
Olanzapine (Pamoate)	SGA	15 studies = YES 2 studies = NO	N/A	**17 Studies;** Kaplan et al,2013, Malik et al, 2017, Offord et al,2012, Marcus, et al, 2015, Lytle, et al, 2017,Viala et al,2012 McCreath, et al 2021,Fricchione, et al, 2017, Shaheen et al 2021,Nielsen et al, 2014,Lin et al, 2019, Xue et al,2019,El Khoury et al,2020 ,Pilon et al,2017, Chan et al,2021,, Druais et al,2016, Lee et al, 2021,	Participants taking SGA LAI's gained weight especially with Olanzapine LAI. No statistical or major clinical differences noted between SGA and FGA. FGA may have cost benefit advantages.
Zuclopenthixol	FGA	2 studies = YES 1 study = NO	N/A	**3 Studies;** Fricchione, et al, 2017, Orietta et al, 2018,Nielsen et al, 2014	SGA LAI patients experienced weight gain with Olanzapine LAI.

Figure 1 : Data Sources Utilized

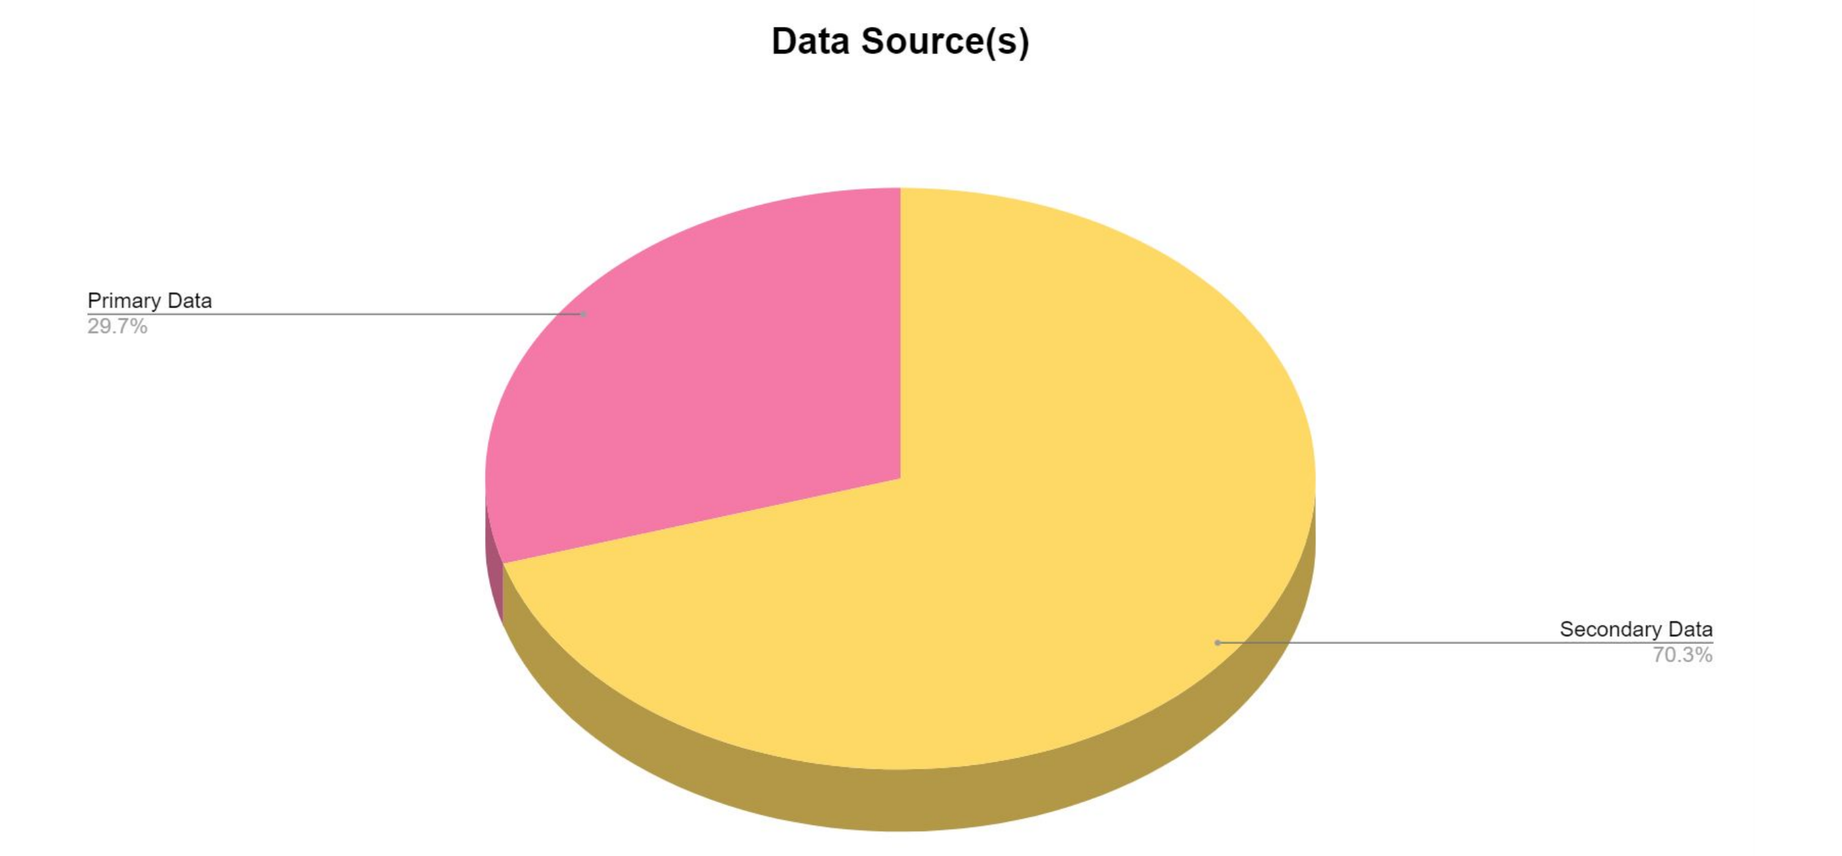
Data Source(s)
Primary Data
29.7%
Secondary Data
70.3%

Figure 2 Adherence in Schizophrenia Studies

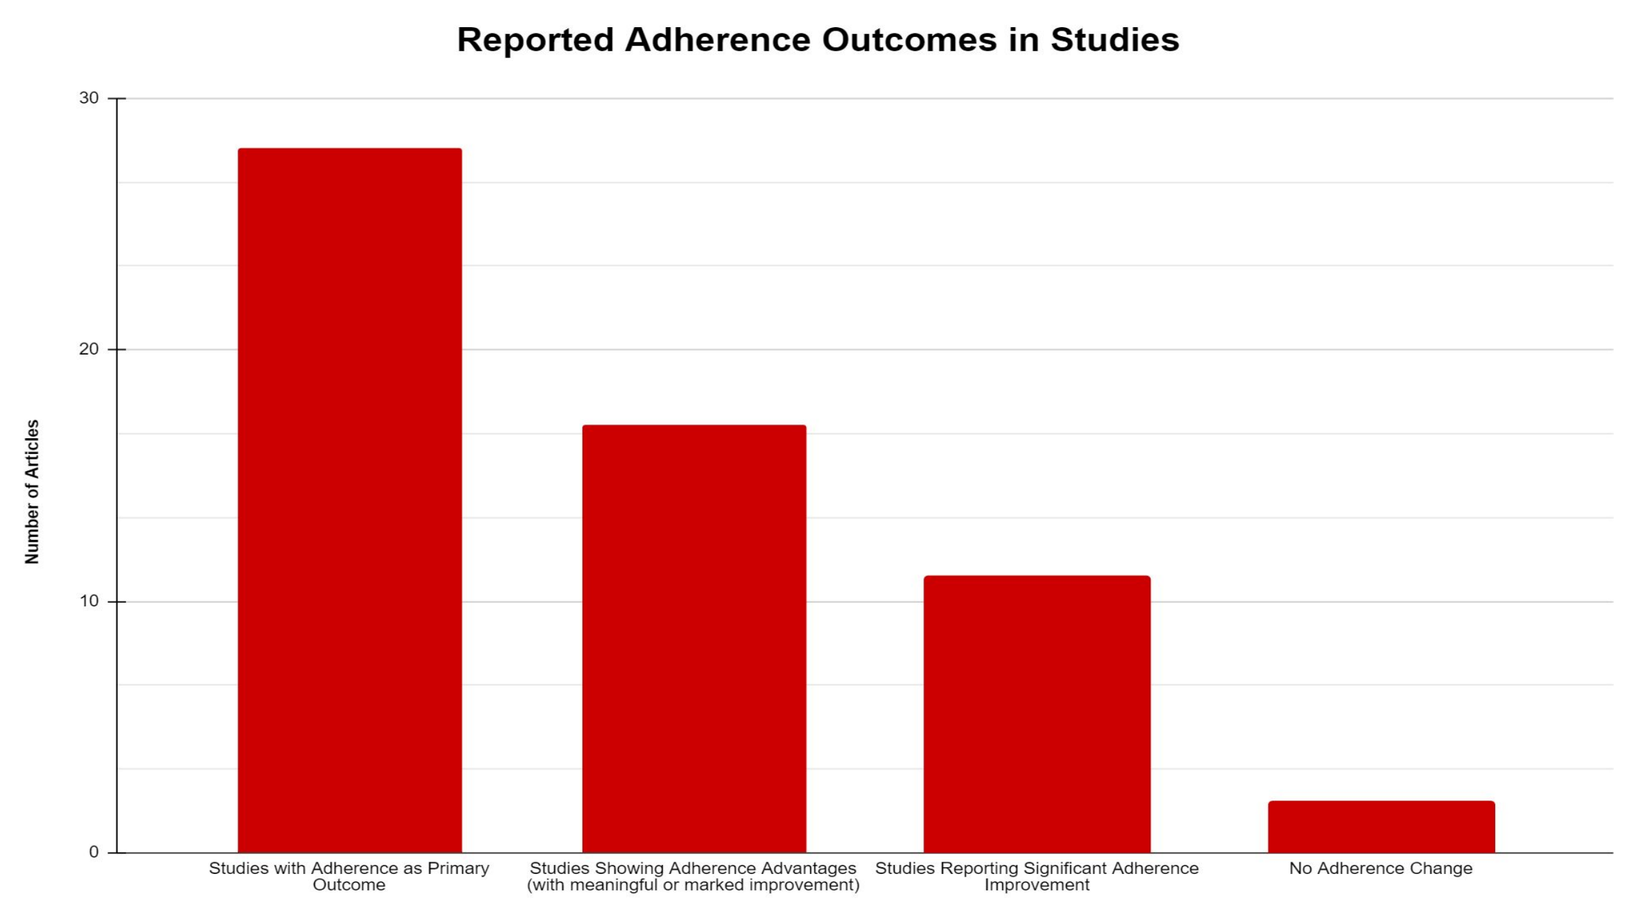
Reported Adherence Outcomes in Studies
Number of Articles
30
20
10
0
Studies with Adherence as Primary Outcome
Studies Showing Adherence Advantages (with meaningful or marked improvement)
Studies Reporting Significant Adherence Improvement
No Adherence Change

Figure 3: Psychiatric Hospitalization, Rehospitalization and Cost Benefits

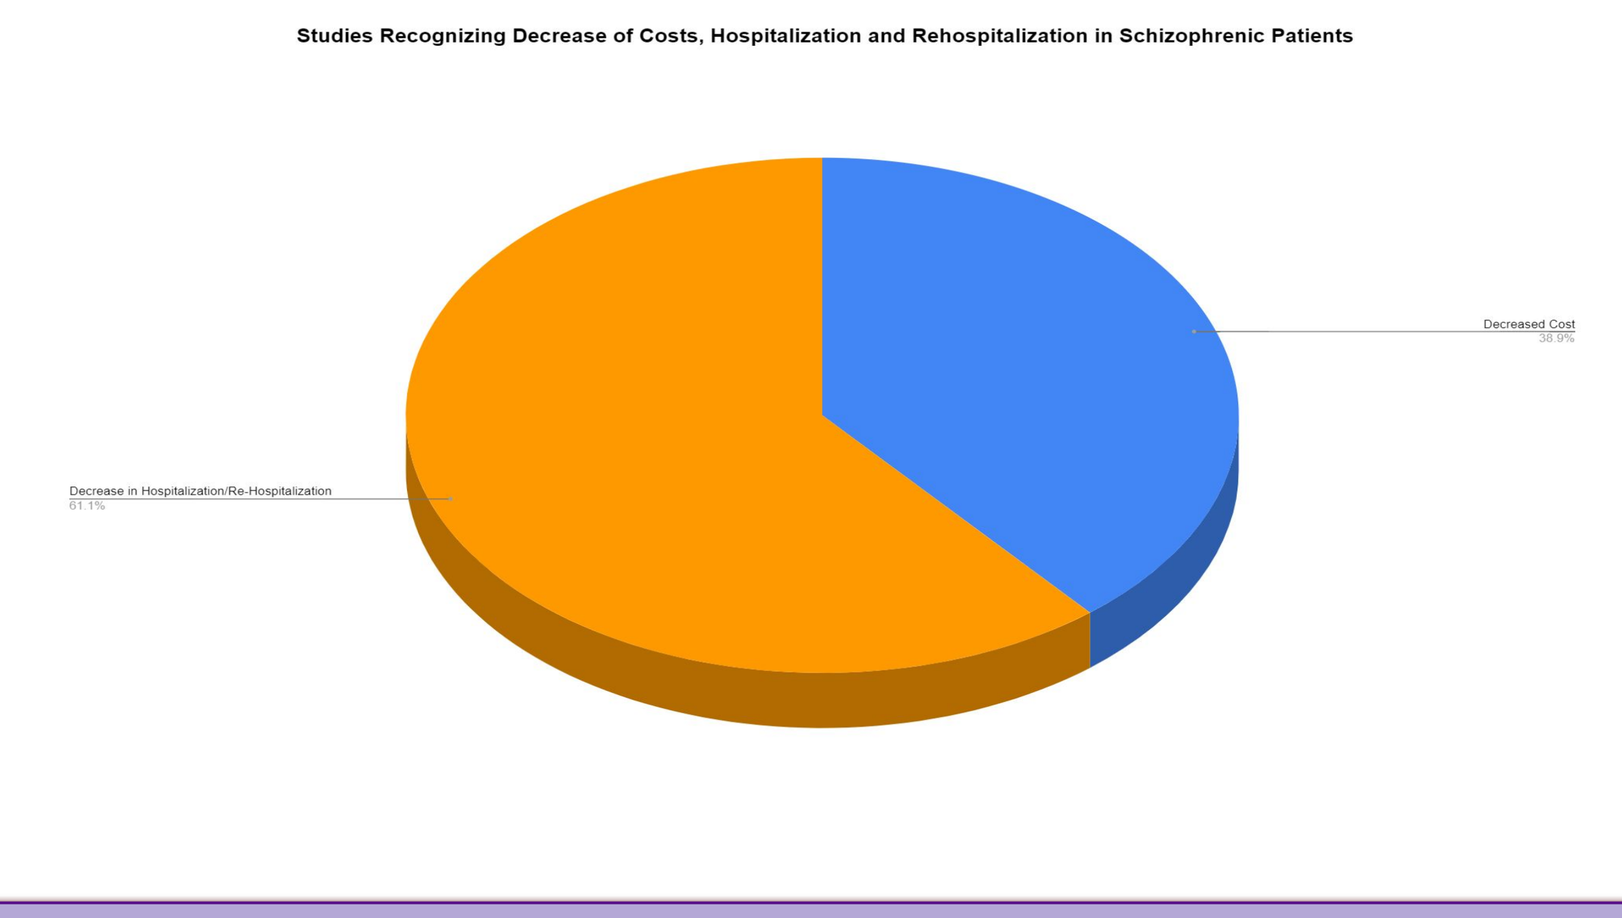
Studies Recognizing Decrease of Costs, Hospitalization and Rehospitalization in Schizophrenic Patients
Decreased Cost
Decrease in Hospitalization/Re-Hospitalization

Figure 4:: First-Time Use vs Maintenance Use

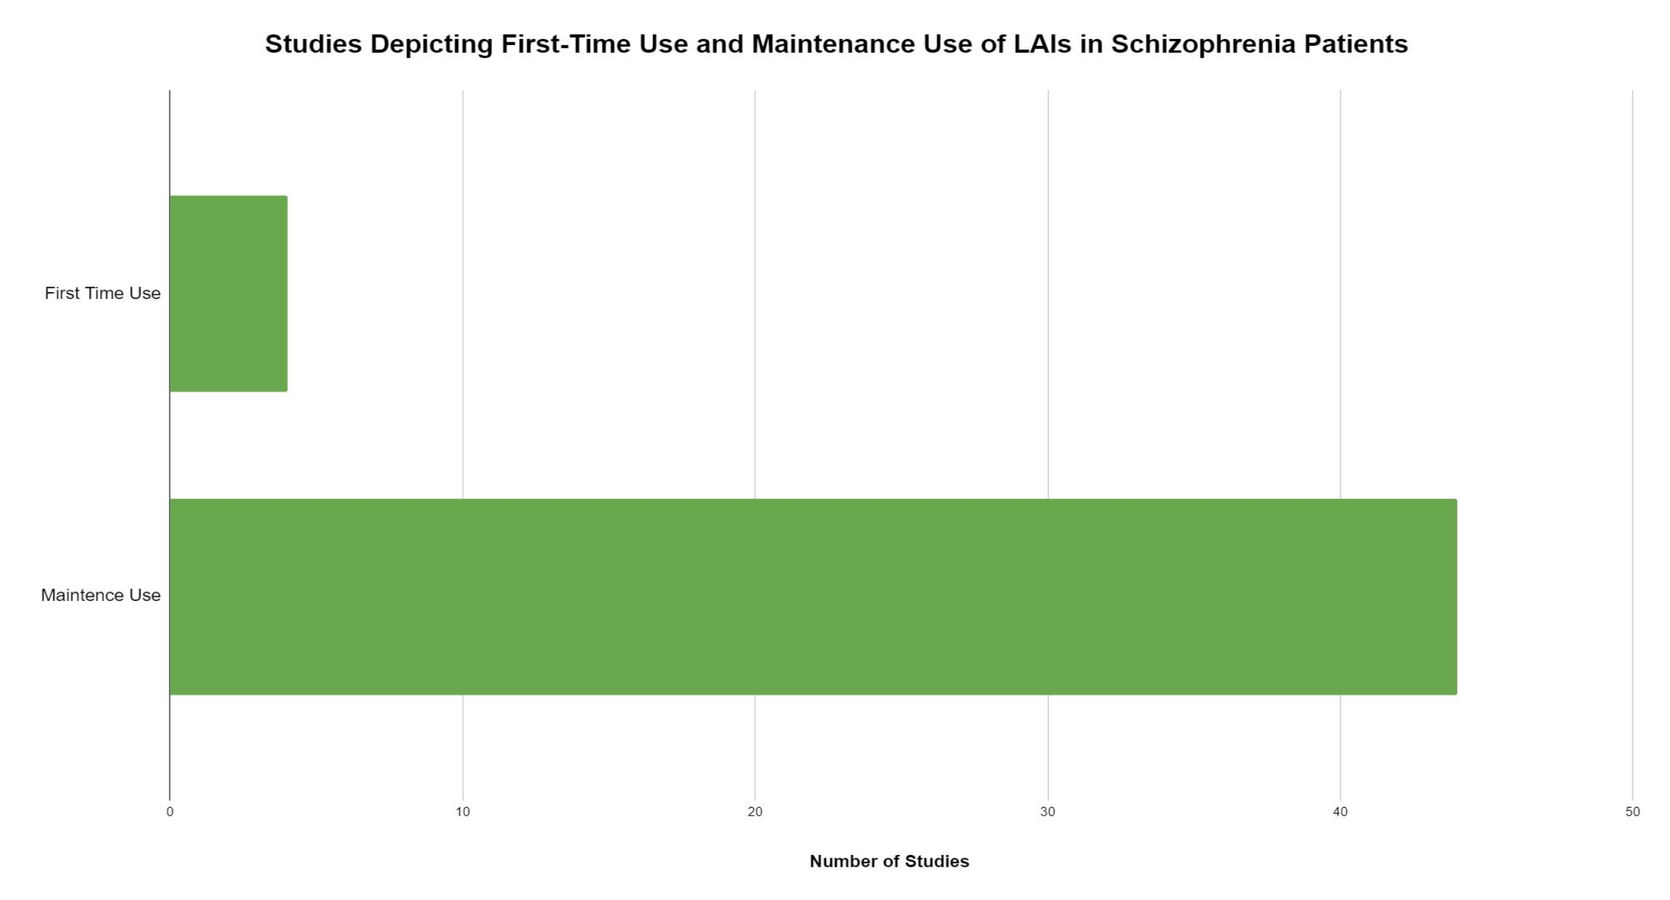
Studies Depicting First-Time Use and Maintenance Use of LAIs in Schizophrenia Patients
First Time Use
Maintence Use
0
10
20
30
40
50
Number of Studies

DISCUSSION

- Patients receiving newer generation LAIs appear to demonstrate better tolerability, better odds of nonadherence, discontinuation rates, and hospitalization, although the reviewed evidence is mostly inconclusive one way or another.
- Evidence also indicates substantial economic advantages and cost savings with newer LAI generation, and over their oral counterparts.
- Evidence regarding superiority of LAIs (over conventional treatment, or over one another) with respect to duration and psychiatric hospitalization appears to be limited and somewhat contradictory.
- Some specific product types (for example, Paliperidone) appear to perform better regarding adherence compared to other products in the same class, while no one particular product has clear superiority over others with respect to a range of outcomes studies.
- Studies investigating the impact of patient characteristics and demographics on adherence and psychiatric hospitalization are extremely limited, and the role of such factors in LAI effectiveness is not well understood.
- While efficacy advantages of LAI and adherence improvements are well documented, RWE regarding risk-benefits in terms of toxicity, side effects and ADRs is beginnig to emerge.

CONCLUSION

Newer generation LAIs appear to be mostly superior compared to the older LAIs, but LAIs overall demonstrate a clear superiority over their oral counterparts with respect to adherence advantages and psychiatric hospitalization. More RWD from RCTs and observational studies are needed than is reported in the literature. Although not conclusive or adequate, evidence mostlysupports LAI's positive impact on adherence in maintenance treatment, health outcomes, and hospitalization. Some evidence documenting the effectiveness of early-use LAIs is beginning to emerge, but uncertainties remain regarding the effectiveness of newer LAIs over first-generation LAIs.

DISCLOSURE

Authors have NO financial disclosure or conflicts of interest with the presented research in this presentation.