Poster CO90

Aditi Kataria | aditi.kataria@novartis.com
Evelina Jaegerskog | evelina.jaegerskog@novartis.com
Ramandeep Jindal | ramandeep.jindal@novartis.com
Xiting Cao | xiting.cao@novartis.com
Amit Ahuja | iamitahuja@gmail.com
Ricardo Viana | Ricardo.viana@novartis.com

A Systematic Literature Review of the Clinical Burden in Patients with Myelodysplastic Syndromes

Kataria A[1], Jaegerskog E[2], Jindal R[1], **Xiting Cao[3]**, Ahuja A[1], Viana R[3]

[1]Novartis Healthcare. Pvt. Ltd, Hyderabad, Telangana, India
[2]Novartis Sverige AB, Stockholm, Sweden
[3]Novartis Pharmaceuticals, East Hanover, NJ, USA
[4]Novartis Pharma AG, Basel, Switzerland

Scan to obtain:
- Poster

https://http://novartis.medicalcongressposters.com/Default.aspx?doc=aaf51

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

LIMITATIONS & CONCLUSIONS

- Most of the data was from the US (n=36) and more studies are needed from across geographies to draw more meaningful conclusions.
- Majority of the studies included in the present review were retrospective and thus, associations with respect to various prognostic factors could be speculative.
- There is a substantial clinical burden associated with MDS; age, gender, cytogenetics, comorbidities and blood transfusion being the predominant factors that predict survival and QoL.
- Lower survival rates observed among higher-risk patients compared to lower-risk patients despite increased usage of HMAs treatment, suggest need for more effective therapies, especially in the higher-risk patients.
- Anemia, thrombocytopenia and neutropenia contributed major symptom burden among MDS patients.
- Comorbidities were found associated with lower QoL and consequently reduced OS among MDS patients.

This study was funded by Novartis Pharma AG. Basel, Switzerland.
Poster presented at ISPOR US 2022, xxxx, xxxxxxx, 22 April 2022.

BACKGROUND AND OBJECTIVE

- The myelodysplastic syndromes (MDS) are a group of disorders characterized by inefficient hematopoiesis leading to cytopenia, with a high-risk of progression to acute myeloid leukemia (AML).[1]
- MDS presents a diverse etiology through epigenetic factors, ethnic differences, age and gender related factors, making diagnosis and treatment challenging.[1,2]
- GlobalData estimates an annual increase in MDS cases by 2.40% from 90,000 in 2018 to 112,000 cases in 2028 in all eight major geographies (the United States, France, Germany, Italy, Spain, the United Kingdom, Japan and Canada).[3]
- The revised International Prognostic Scoring System (IPSS-R) is currently used for estimating prognosis and classifying patients with MDS into five categories: very low, low, intermediate, high and very high-risk. Physicians treat MDS patients based on the broader categories of lower-risk (very low, low, intermediate) and higher-risk (very high, high, intermediate).[4,5]
- Treatment goals for lower-risk categories include preventing complications and reducing transfusion burden.[1,2]
- Patients with higher-risk MDS require more aggressive treatment, which may include hypomethylating agents (HMAs) and high-dose chemotherapy. Despite existing treatments, patients with higher-risk MDS have a lower overall survival and a higher rate of progression to AML, suggesting an unmet need for safe and effective treatments in the management of MDS.[4]
- Thus, to better understand the disease heterogeneity , we conducted a systematic literature review (SLR) aimed to identify and summarize the clinical burden in patients with MDS.

METHODS

- A comprehensive literature search was conducted from Jan 2011 to April 2021 across the (*Excerpta Medica* database [EMBASE®], MEDLINE and MEDLINE In process database).
- The literature search results were screened according to the pre-defined inclusion criteria, and included MDS patients irrespective, of risk categories, and were screened, first by title and abstract, and then by full-text.
- The SLR was conducted using a three-review process i.e screening to retrieve relevant studies and data extractions into pre-defined extraction grids were completed by two reviewers and any discrepancies were resolved by a third independent reviewer.

RESULTS

- A total of 67 observational studies reported data on clinical and humanistic burden of MDS. Most of them were conducted in the US (n=36) or in Europe (n=26).
- Preferred Reporting Items for Systematic Reviews and Meta-Analyses (PRISMA) flow for the SLR is presented in **(Figure 1)**.

Figure 1: Flowchart of the study selection

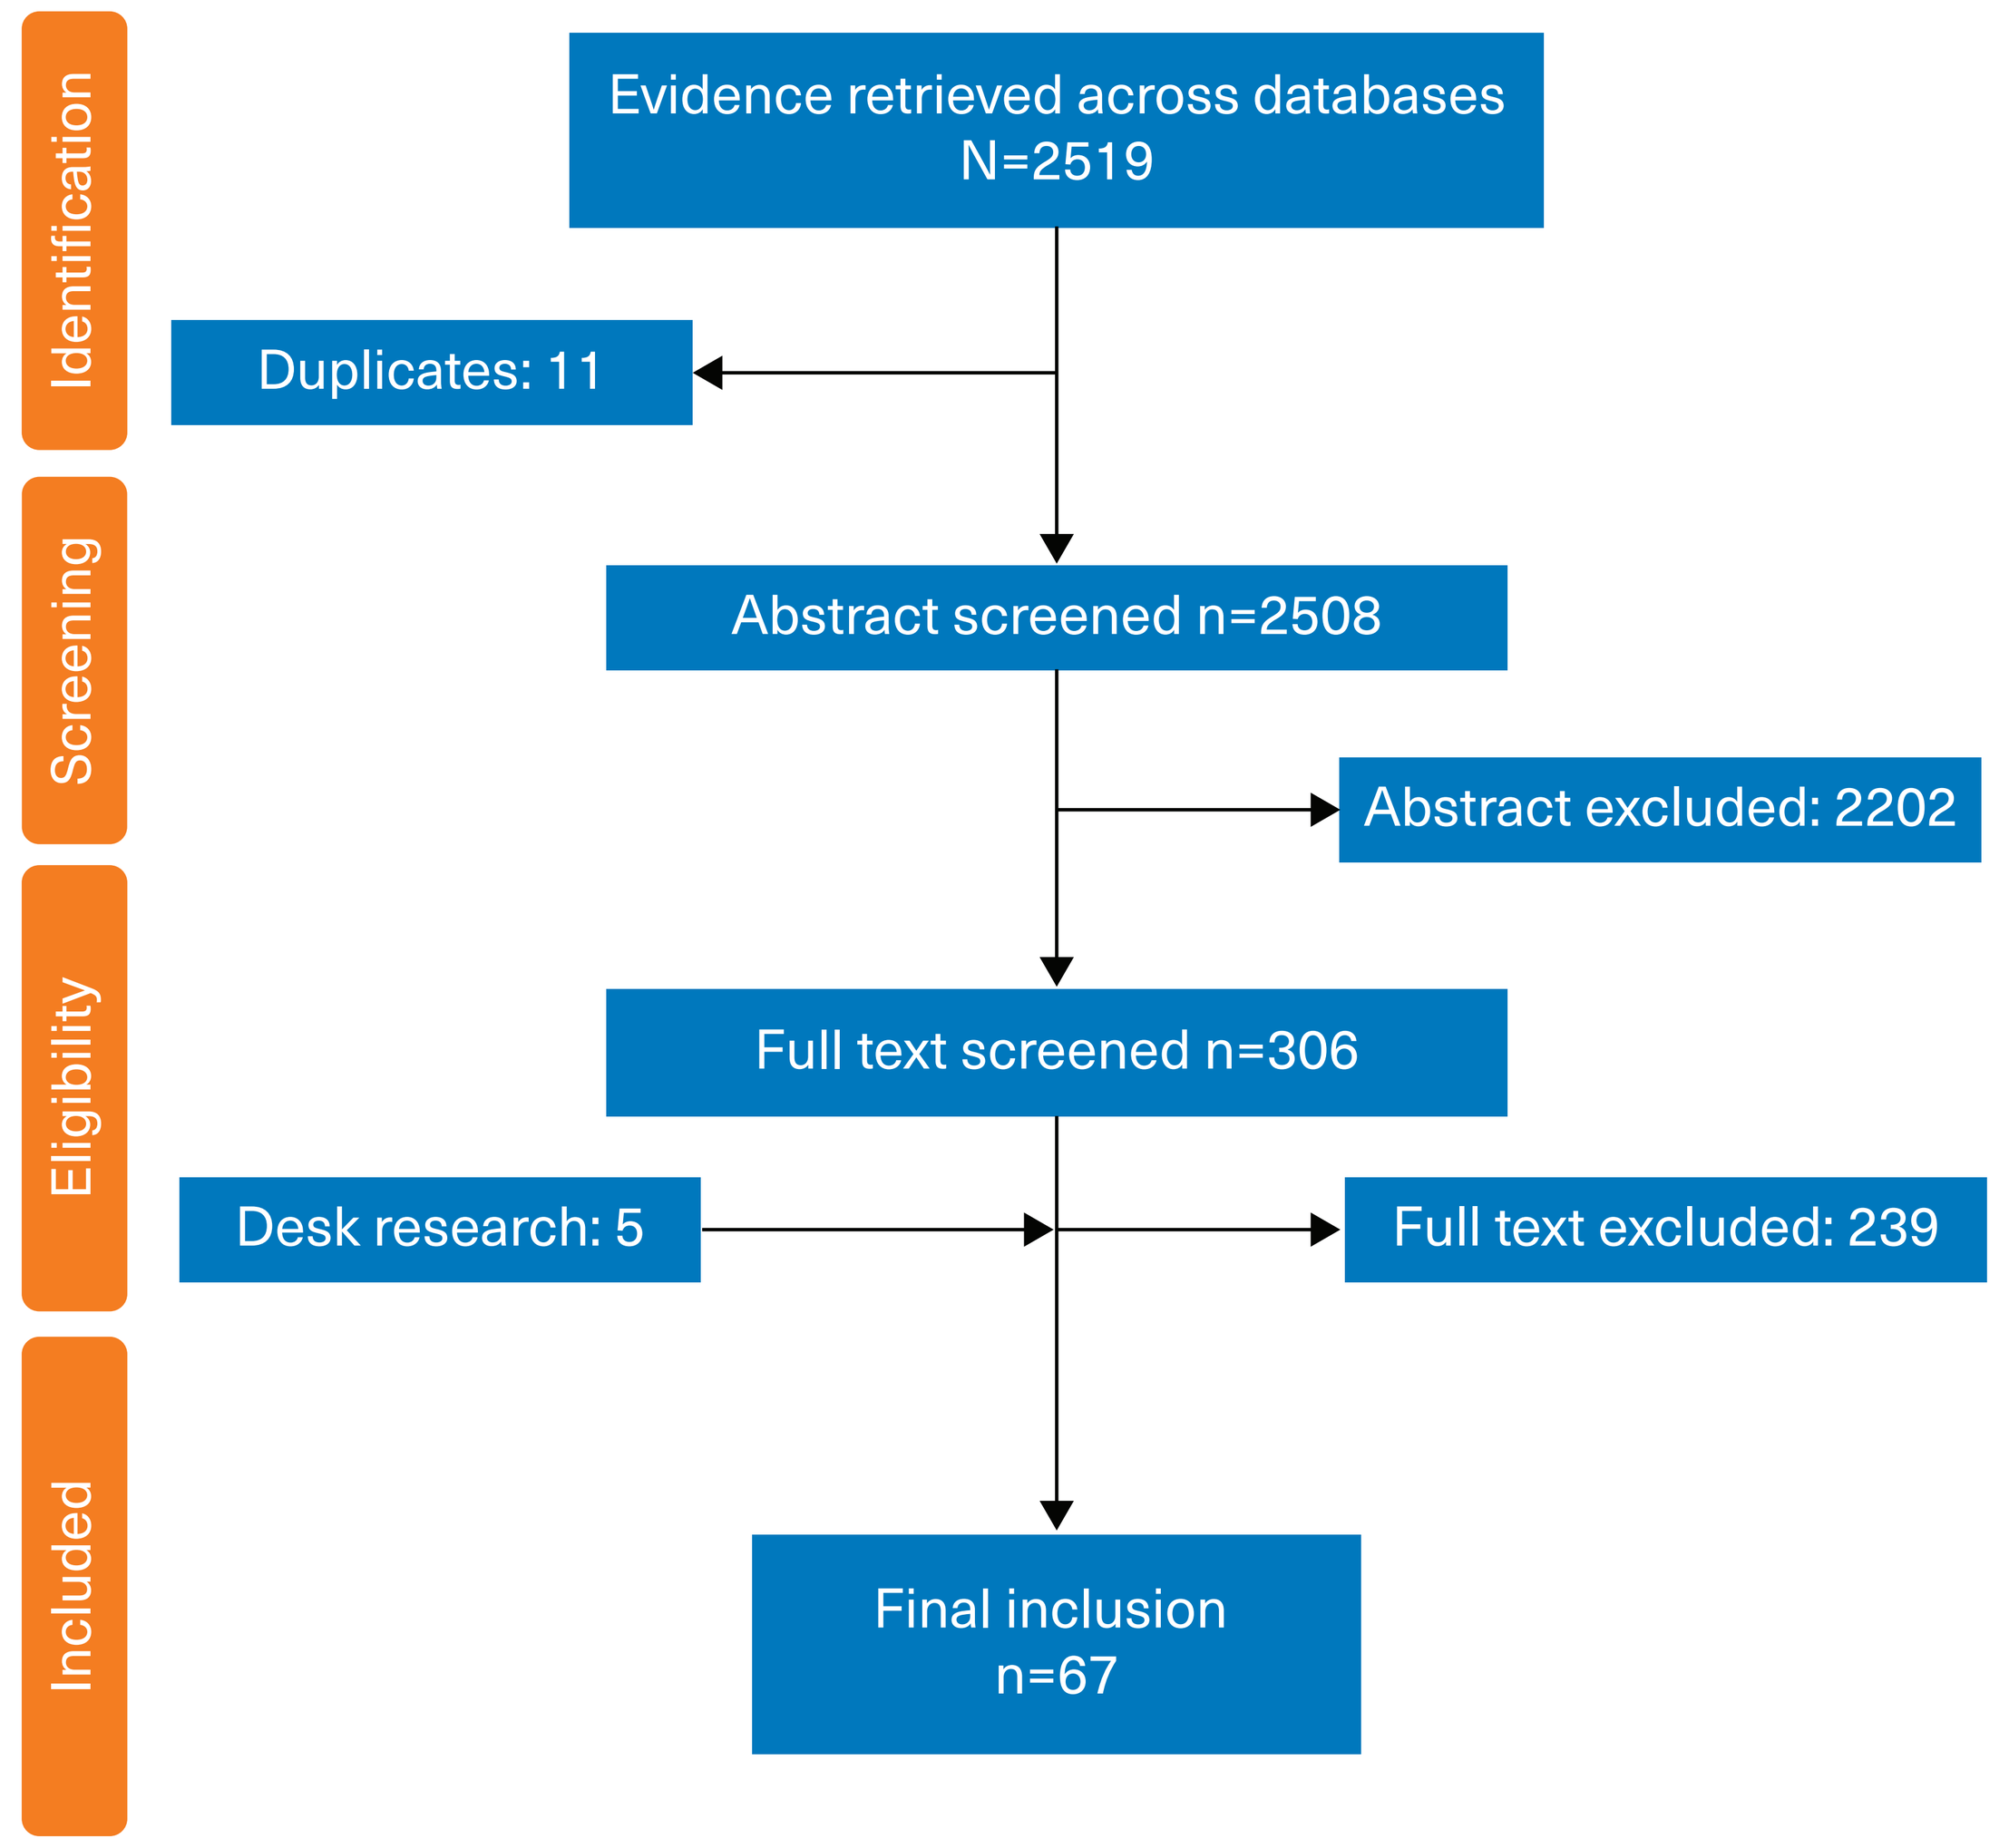

Identification
Screening
Eligibility
Included
Evidence retrieved across databases N=2519
Duplicates: 11
Abstract screened n=2508
Abstract excluded: 2202
Full text screened n=306
Desk research: 5
Full text excluded: 239
Final inclusion n=67

- The mean age of the patients ranged from 61 years to 79 years, with over 50% of patients in a majority of studies (n=46) being males.
- Data from six studies indicated that stem cell transplantation was administered to 7% to 11% of patients. The mean percentage of patients requiring transfusions were 56.3% (14.5% to 100%) with data available from 21 studies.

Survival

- Overall, 54 studies reported data on survival across MDS risk categories.
- High-risk MDS patients were four times less likely to survive as compared to low-risk MDS patients over 85 months (HR, 4.50, 2.80-7.10, p<0.001).
 - Among the studies reporting data across all MDS risk groups, median survival was the highest in very low-risk MDS group (ranging from 60 months to 136 months), followed by low-risk MDS group (ranging from 40 to 90 months), intermediate risk MDS group (ranging from 25 to 35 months), high-risk MDS group (ranging from 15 to 30 months), and very high-risk MDS group (ranging from 6 to 15 months). **(Figure 2a)** and **(Figure 2b)**
 - The data shows that there is a slight overlap of the OS between intermediate and high-risk patients with a difference of only a 5 to 10 month. This implies that patients with intermediate-risk MDS were more prone to develop high-risk MDS as opposed to low-risk MDS.
- Patients with high-risk MDS had a lower OS despite an increased usage of HMAs treatment from 3.6% to 43%.
- Presence of comorbidities (two times), karyotyping (2–5 times), and transfusion dependency were predictive of low survival among MDS patients, with effects being more prominent in the high-risk MDS patients.[6]

Figure 2a: Median survival across IPSS-R risk groups, in months

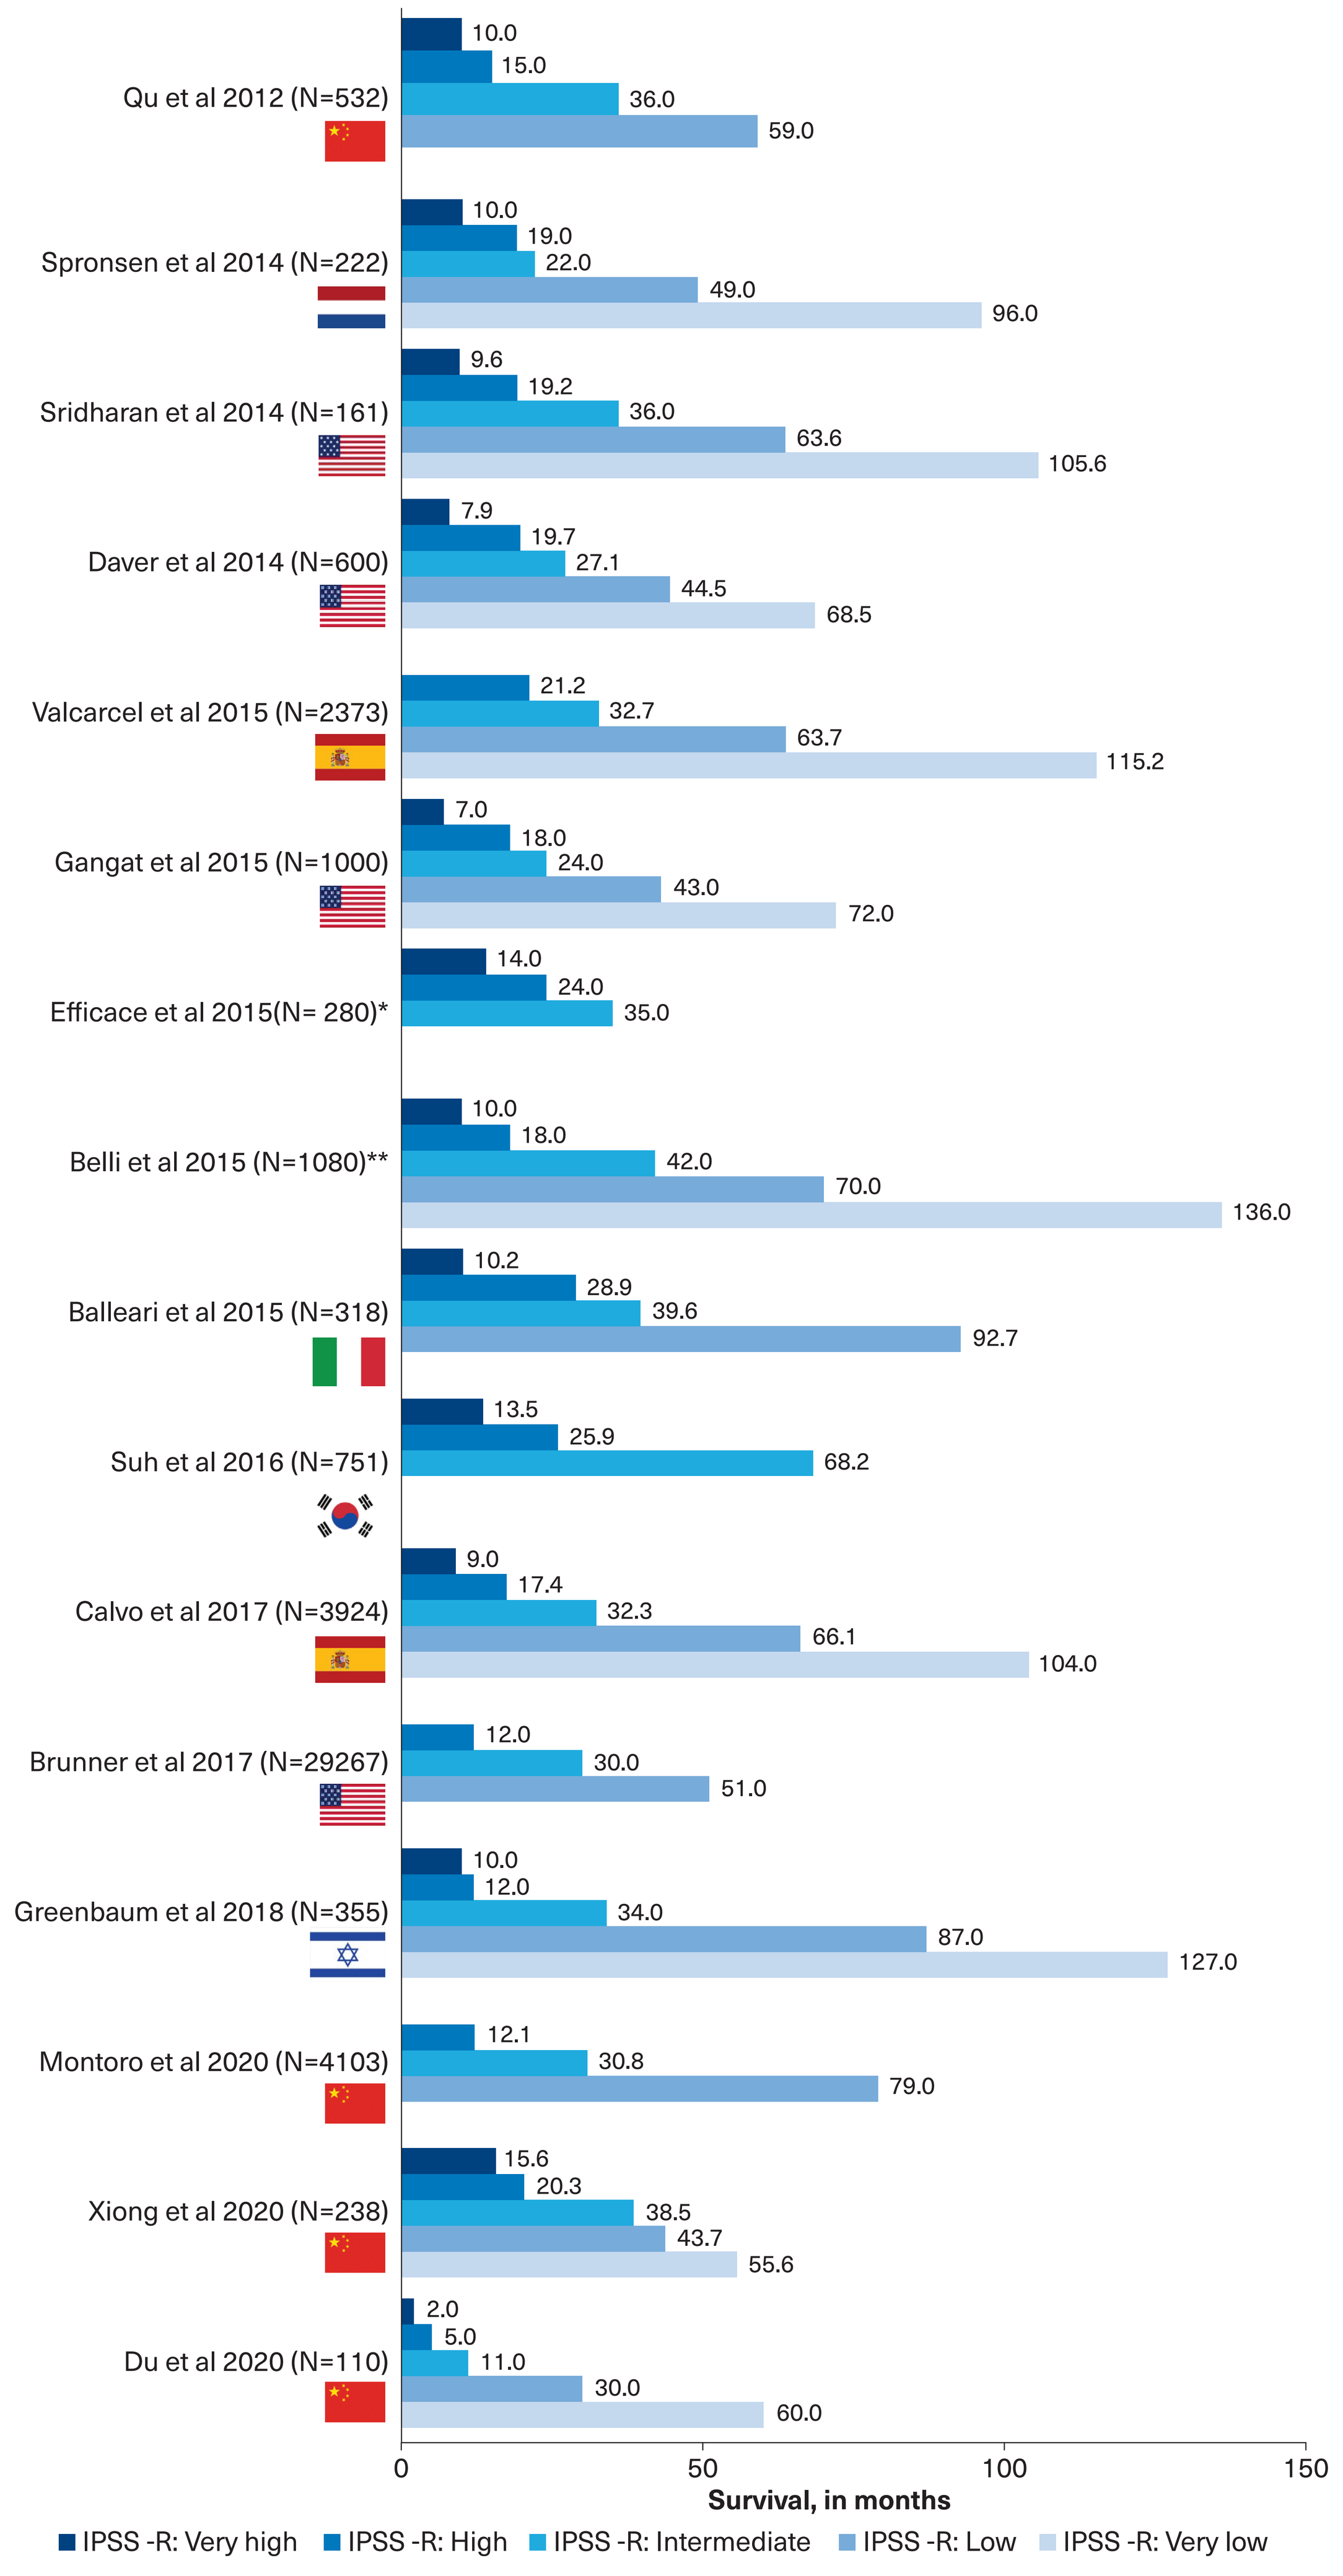

Qu et al 2012 (N=532)
10.0
15.0
36.0
59.0
Spronsen et al 2014 (N=222)
10.0
19.0
22.0
49.0
96.0
Sridharan et al 2014 (N=161)
9.6
19.2
36.0
63.6
105.6
Daver et al 2014 (N=600)
7.9
19.7
27.1
44.5
68.5
Valcarcel et al 2015 (N=2373)
21.2
32.7
63.7
115.2
Gangat et al 2015 (N=1000)
7.0
18.0
24.0
43.0
72.0
Efficace et al 2015(N= 280)*
14.0
24.0
35.0
Belli et al 2015 (N=1080)**
10.0
18.0
42.0
70.0
136.0
Balleari et al 2015 (N=318)
10.2
28.9
39.6
92.7
Suh et al 2016 (N=751)
13.5
25.9
68.2
Calvo et al 2017 (N=3924)
9.0
17.4
32.3
66.1
104.0
Brunner et al 2017 (N=29267)
12.0
30.0
51.0
Greenbaum et al 2018 (N=355)
10.0
12.0
34.0
87.0
127.0
Montoro et al 2020 (N=4103)
12.1
30.8
79.0
Xiong et al 2020 (N=238)
15.6
20.3
38.5
43.7
55.6
Du et al 2020 (N=110)
2.0
5.0
11.0
30.0
60.0
0
50
100
150
Survival, in months
IPSS -R: Very high
IPSS -R: High
IPSS -R: Intermediate
IPSS -R: Low
IPSS -R: Very low

IPSS-R: Revised International Prgnostic Scoring System; *Global study;**South American countries (Argentina, Brazil, Chile)*

Figure 2b: Median survival across IPSS risk groups, in months

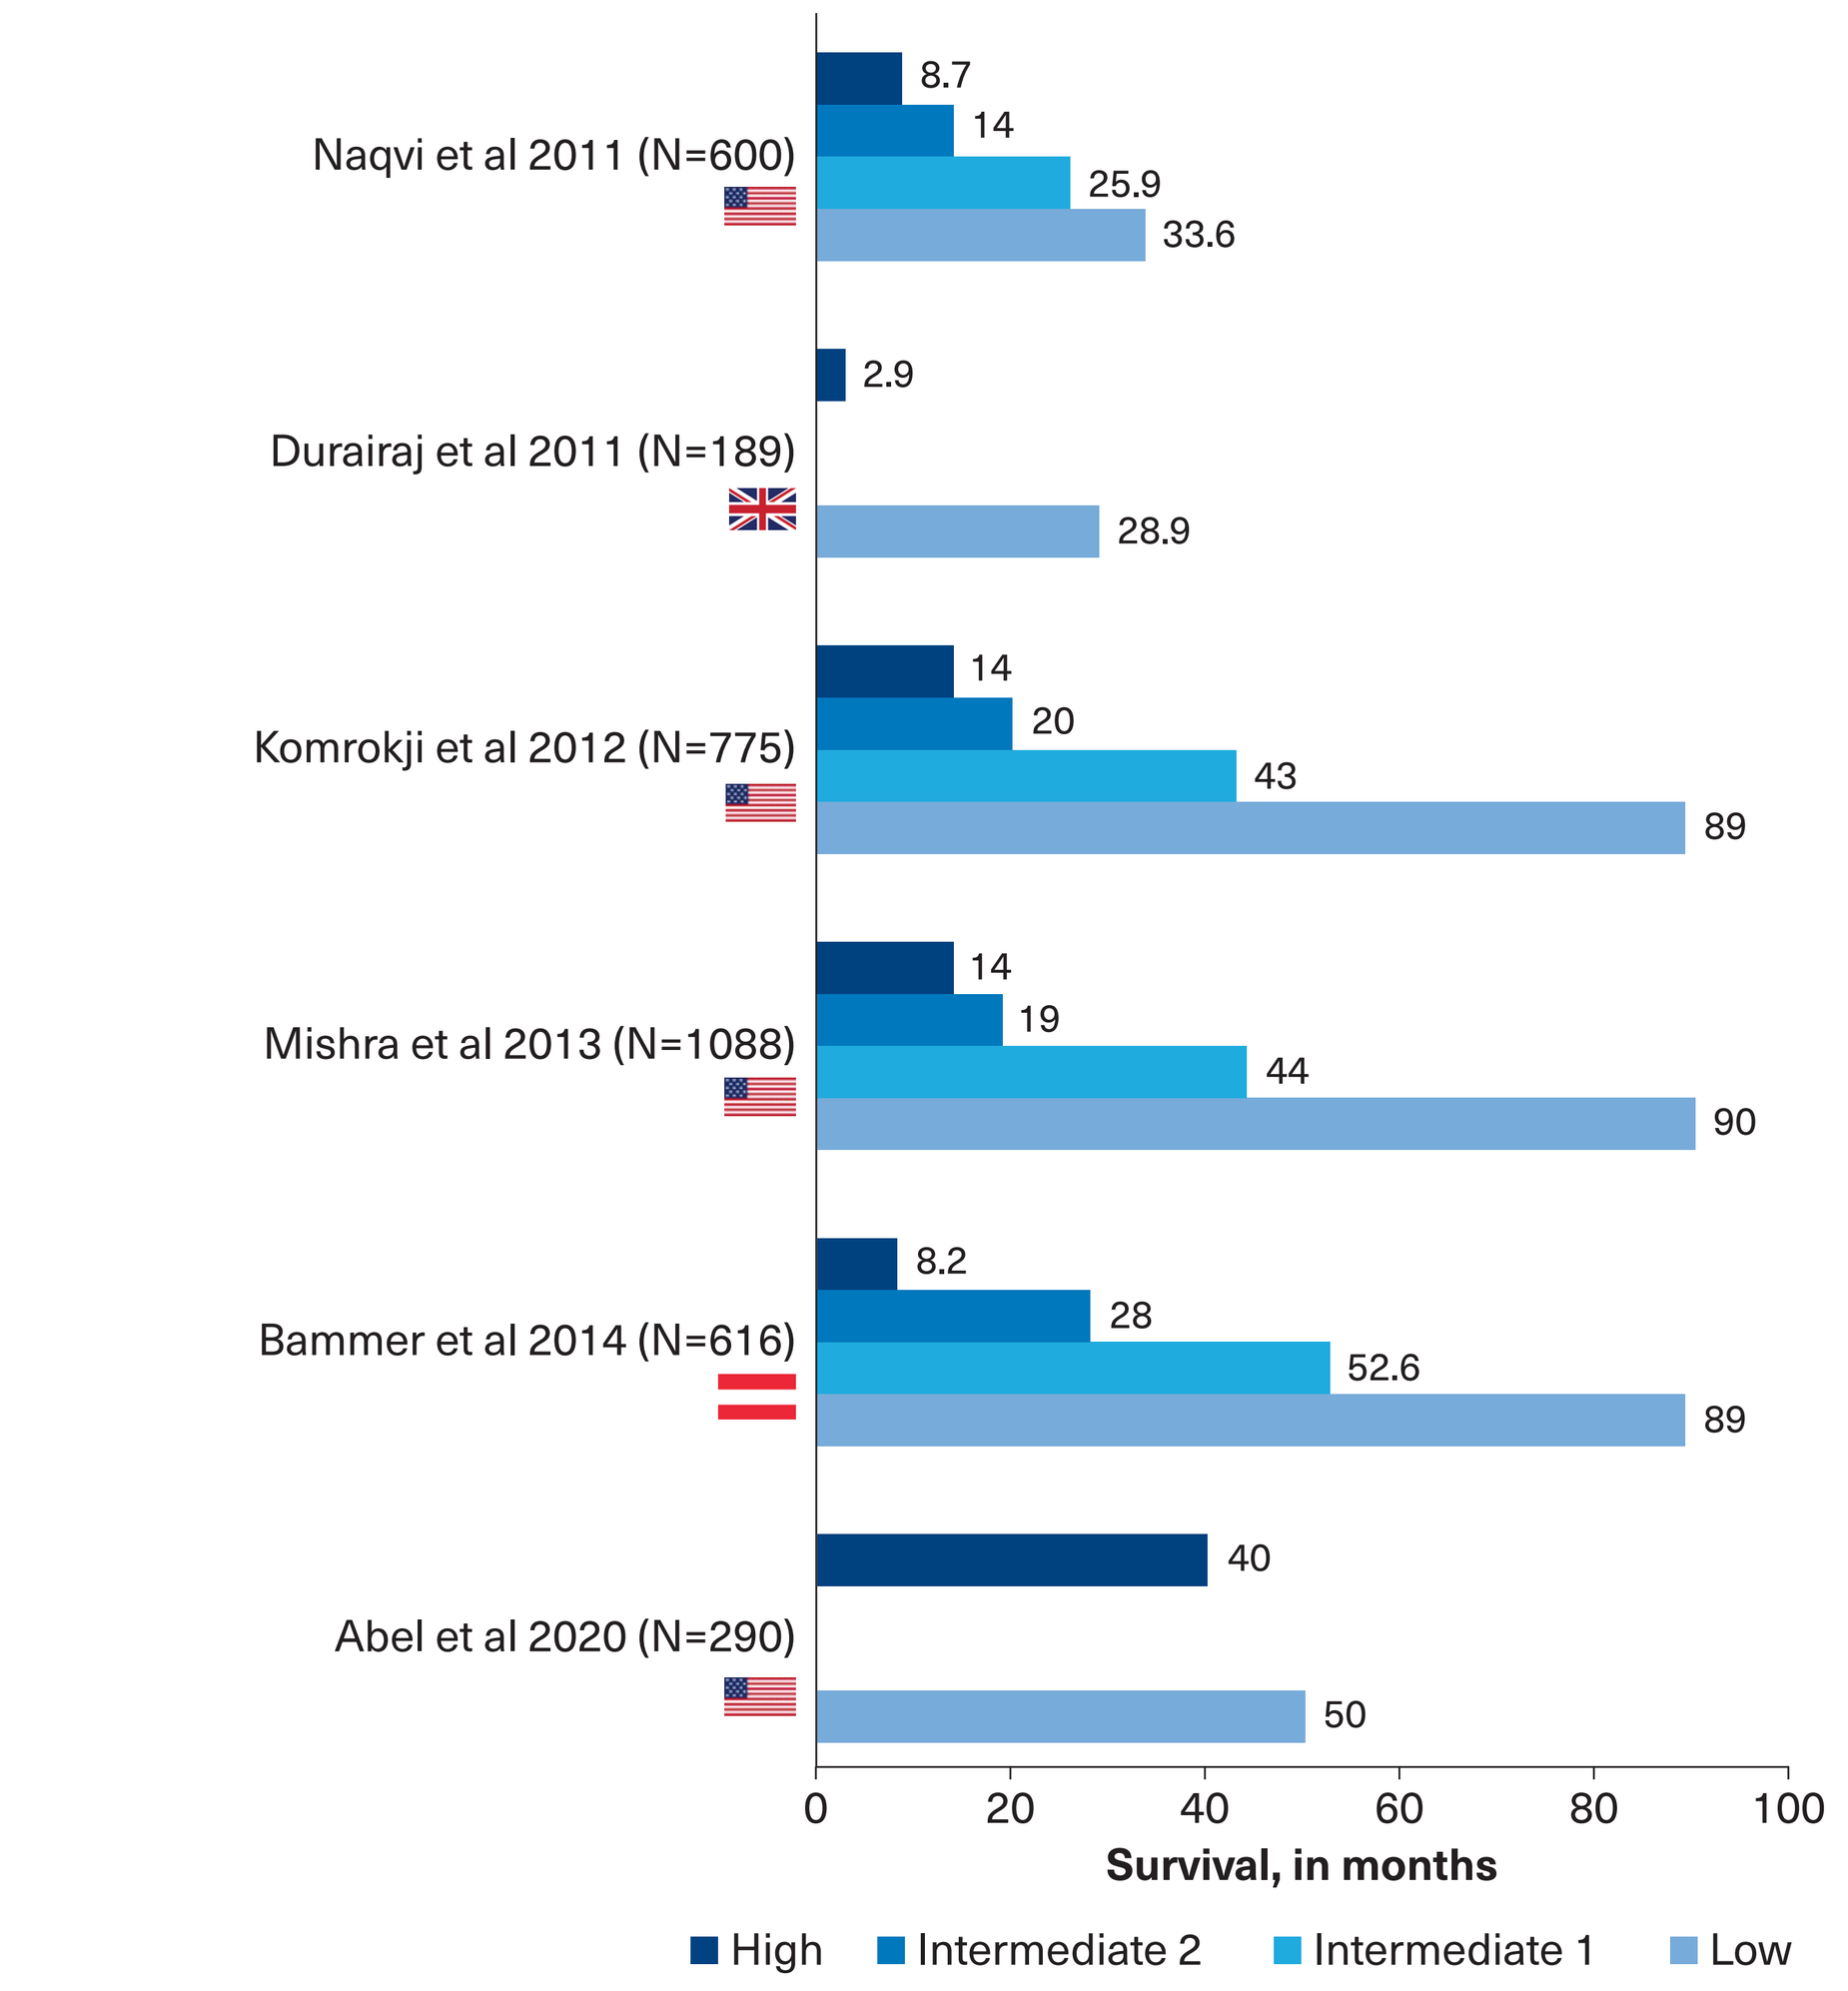

Naqvi et al 2011 (N=600)
8.7
14
25.9
33.6
Durairaj et al 2011 (N=189)
2.9
28.9
Komrokji et al 2012 (N=775)
14
20
43
89
Mishra et al 2013 (N=1088)
14
19
44
90
Bammer et al 2014 (N=616)
8.2
28
52.6
89
Abel et al 2020 (N=290)
40
50
0
20
40
60
80
100
Survival, in months
High
Intermediate 2
Intermediate 1
Low

IPSS: International Prognostic Scoring System

AML transformation rate

- A total of six studies reported data on MDS patients who transformed to AML.
- Progression to AML was a time-dependent variable and significantly associated with survival [HR, 1.80 (1.27-2.55), p<0·001].
- Higher-risk patients showed a higher likelihood of AML transformation in three of six studies as compared to lower-risk patients.
- The rate of AML transformation increased by 1.5 fold as the number of years on MDS increased (55% and 33% in high-risk MDS and intermediate risk MDS patients in the 1st year versus 39% and 19% in the second year, respectively).
- Furthermore, patients with intermediate risk MDS group had a 3 years cumulative incidence of AML evolution [20.70% (95% CI, 17.70-24.30)] along with an OS, analogous to MDS with high-risk MDS patients.
- Patients with karyotype der(1;7) were associated with low AML transformation rates (11%) compared to del (7q) (42%; HR, 3.90 (1.20-12.30), p=0.02) and monosomy 7 (41%; HR, 5.90 (2.50-14.00), p<0.001).

Comorbidities:

- A total of 12 studies reported data on comorbidities.
- Male gender and age (>65 years) were associated with higher mean number of comorbidities (p<0.05) and, consequently, with a poorer OS.
- Diabetes (23%) and cardiovascular disorders (16%) were found to be the most prevalent comorbidities among patients with high-risk MDS.
- Further, cardiovascular disorders (HR, 3.57 p<0.001) were associated with significantly higher-risk of OS among other diseases (hepatic disorders: HR, 2.55, p=0.01, pulmonary disorders: HR, 2.44, p<0.005, renal disease: HR, 1.97, p=0.04).[7]
- Also, presence of comorbidity was associated with poor OS, as the mean Adult comorbidity Evaluation (ACE)-27, Charlson Comorbidity Index (CCI), and hematopoietic cell transplantation comorbidity index (HCT-CI) scores increases, survival decreases. **(Table 1)**

Table 1: Association of comorbidities with mortality/survival

Study	Country	Mortality (Hazard ratios)	Median survival (months)
Rozema et al 2021 (N=291)	Netherlands	CCI score 1: 1.0 (0.7-1.5) CCI score 2-3: 1.3 (0.9-1.9) CCI score ≥4: 2.3 (1.6-3.4)	–
Bammer et al 2014 (N=616)	Austria	HCT-CI: 1.3 (1.1–1.5), p<0.005	–
Daver et al 2014 (N=600)	USA	–	ACE- 27 score None, 0: 45.8 (27.3–64.3) ACE- 27 score Mild, 1: 21.8 (16.6–27.0) ACE- 27 score Moderate, 2: 17.4 (10.5–24.3) ACE- 27 score Severe, 3: 10.0 (6.6–13.4)

ACE: Adult Comorbidity Evaluation; CCI: Charlson Comorbidity Index; HCT-CI: Hematopoietic cell transplantation comorbidity index

Symptom burden and humanistic burden:

- In total, seven studies reported data on symptom burden and fatigue.
- Anemia, thrombocytopenia and neutropenia constituted major symptom burden in more than 50% of MDS patients.
- Percentage of MDS patients with anemia was higher in del(5q) patients (69%), compared to patients with refractory cytopenia with unilineage dysplasia (RCUD), refractory anaemia with ringed sideroblasts (RARS), and refractory cytopenia with multilineage dysplasia (RCMD) (57%, 63% and 64%, respectively).[8]
- Impact of IPSS-R categories on EuroQoL-5D (EQ-5D) scoring, was only marginal and varied per age, gender, comorbidities and transfusion requirement in studies (p<0.001).
- According to the Functional Assessment of Cancer Therapy Anemia (FACT-An) questionnaire, blood transfusions reduced anemia and fatigue in high-risk MDS patients (p=0.016).
- Moreover, a positive correlation was also found between the FACT-An score and hemoglobin value (rs 0.66, p= 0.02).
- Low quality of life (QoL) was associated with reduced survival among MDS patients. Further, presence of ≥2 comorbidities was associated with worsening of QoL.
- The IPSS-R categories had only a marginal impact on EQ-5D scoring among different risk groups, and it varied according to age, gender, comorbidities, and blood transfusion requirements (p<0.001) significantly impacted QoL **(Figure 4)**.[9]
- In addition to female gender, low hemoglobin levels were independently associated with high fatigue severity (p=0.026).

Figure 4: Humanistic burden as measured using EQ-5D VAS

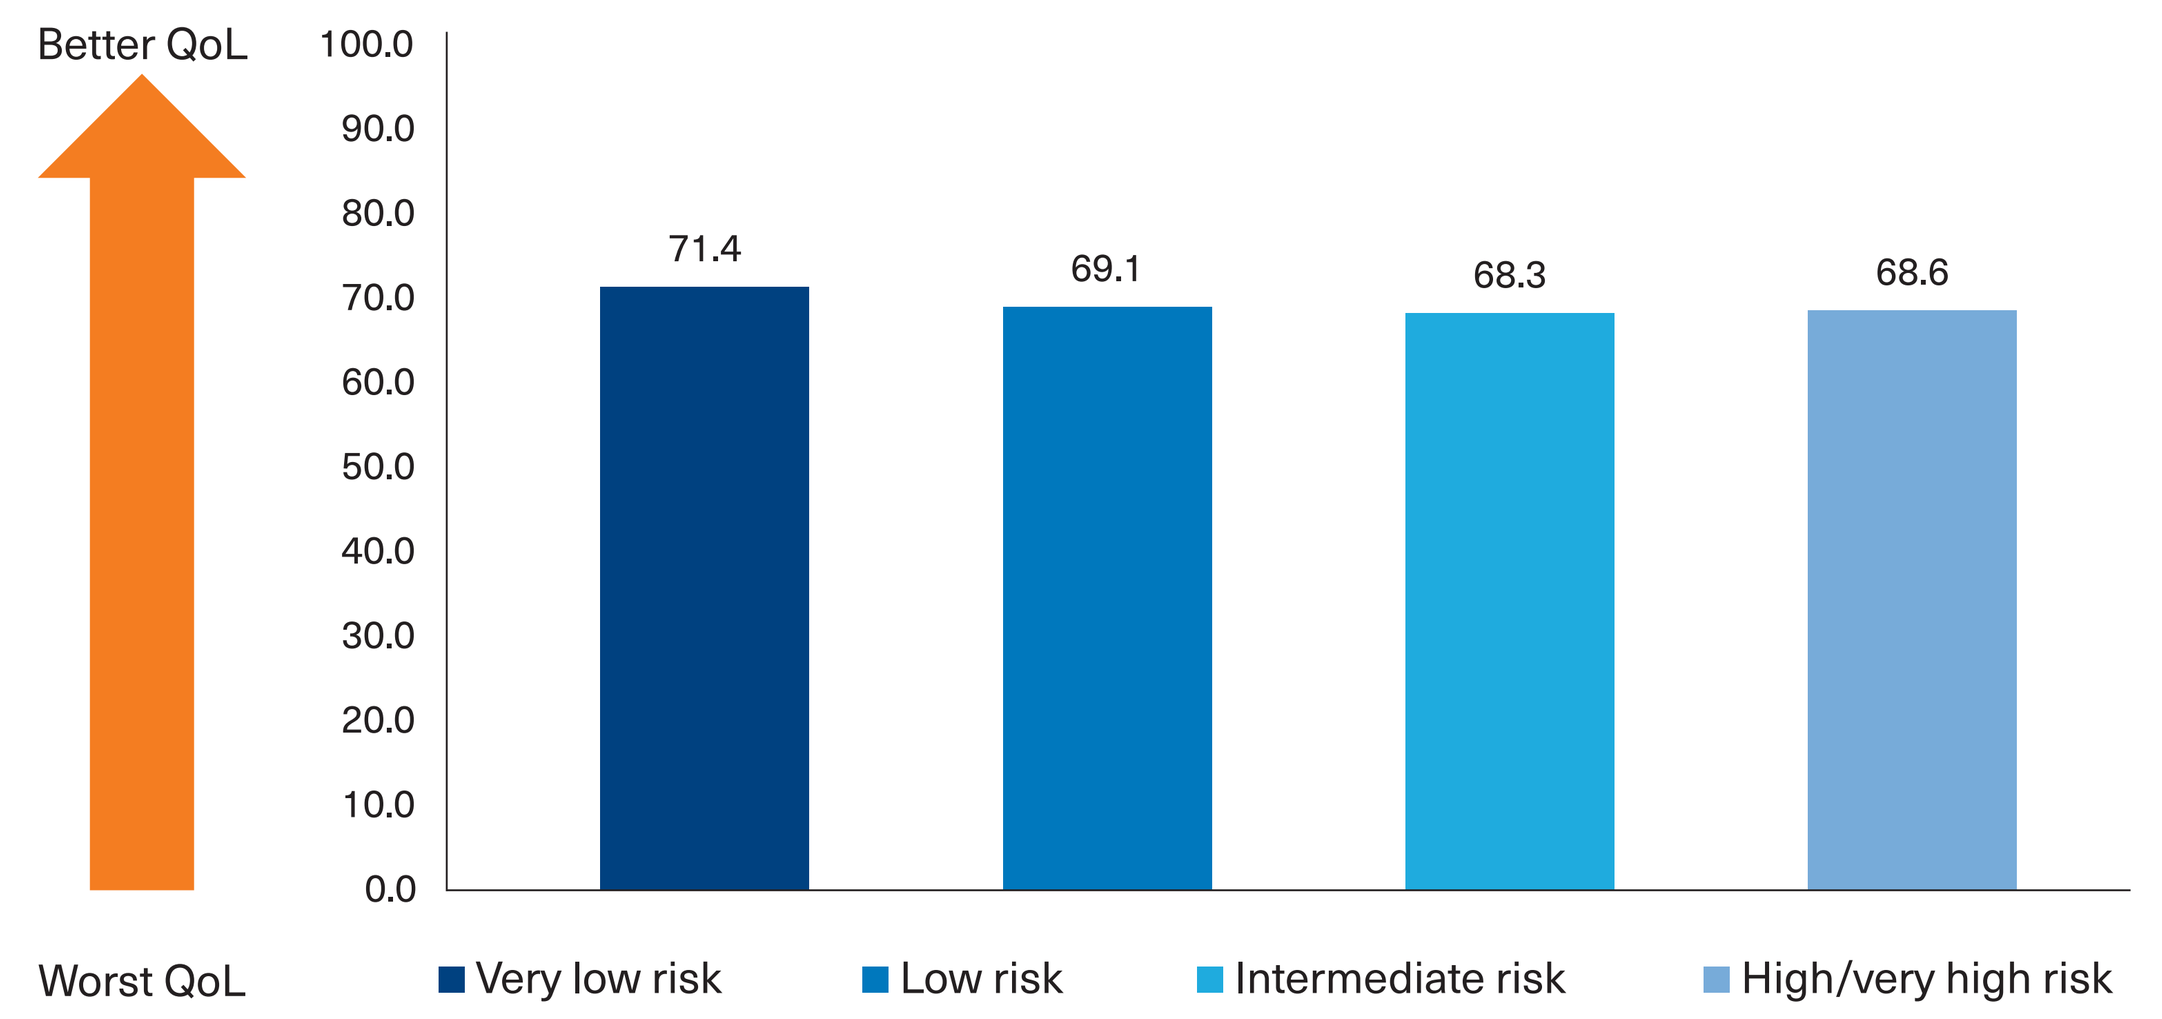

Better QoL
Worst QoL
100.0
90.0
80.0
70.0
60.0
50.0
40.0
30.0
20.0
10.0
0.0
71.4
69.1
68.3
68.6
Very low risk
Low risk
Intermediate risk
High/very high risk

Source: Strauder et al 2018 (N=1985) (Global study)

Acknowledgements

The authors acknowledge Rahul Kumar (Novartis, Hyderabad) for developing the poster and Kuruva Jayaram (Novartis, Hyderabad) for designing the poster layout. The final responsibility for the content lies with the authors.

Conflict of interest

Aditi Kataria, Ramandeep Jindal are employees of Novartis Healthcare Pvt. Ltd., Hyderabad, Telangana, India.
Evelina Jaegerskog is an employee of Novartis Sverige AB and holds stocks. Ricardo Viana is an employee of Novartis Pharma AG, Basel, Switzerland and holds stocks.
Amit Ahuja is a former employee of Novartis Healthcare Pvt. Ltd and was involved in the conduct of this study.
Xiting Cao is an employee of Novartis pharmaceuticals, East Hanover and holds stocks.

References

1. C. M, *The New England journal of medicine*, vol. 383, no. 14, pp. 1358-74, 2020.
2. F. P, *Annals of oncology : official journal of the European Society for Medical Oncology*, vol. 32, no. 2, pp. 142-56, 2021.
3. "Global data," 2021. [Online]. Available: https://wwwglobaldatacom/myelodysplastic-syndrome-diagnosed-incident-cases-set-to-reach-112000-in-2028-says-globaldata/.
4. G. G, *Hematology*, vol. 95, no. 11, pp. 1399-420, 2020.
5. "NCCN guidelines for patients," 2021. [Online]. Available: https://www.nccn.org/.
6. M. L, *Latin-American MDS group - GLAM. EHA Library*, 2018.
7. D. P. MG, *Haematologica*, vol. 96, no. 3, pp. 441-9, 2011.
8. L. M, *British Journal of Haematology*, vol. 170, no. 5, pp. 687-93, 2015.
9. S. R, *Leukemia*, vol. 32, no. 6, pp. 1380-92, 2018.