Direct Cost Components of Lupus Nephritis in Brazilian Private Healthcare: An Expert Panel Approach

Poster No. EE173

Silva, DO[1]; Santana, P[1]; Kano, B[2]; Kashiura, D[2]; David, N[3]; Gazzotti, MR[3]; Bernardino, G[1]

[1]Value Evidence and Outcomes, GlaxoSmithKline, Rio de Janeiro, Brazil
[2]IQVIA Real World Insights, São Paulo, Brazil
[3]Medical Affairs, GlaxoSmithKline, Rio de Janeiro, Brazil

Background

- Systemic lupus erythematosus (SLE) is a chronic inflammatory autoimmune disease that progressively affects several organs, with a usual evolution comprising periods of activity and remission[1]
- When this disease affects the kidneys, the individual can develop lupus nephritis (LN), a type of glomerulonephritis and one of the most serious manifestations of SLE; LN is associated with high morbidity and mortality[1–3]
- It is estimated that 40% of patients with SLE develop LN and 5–20% progress to end-stage kidney disease (ESKD).[1,4] Management of ESKD involves dialysis and evaluation for kidney transplantation[1]
- LN is categorized into different histological classes, specifically classes I–VI, associated with varying prognoses and speed of progression to end-stage kidney disease. Classes III and IV are more strongly associated with irreversible nephron loss[1]
- Patients with LN have greater health resource use and costs compared with those without LN[5]

Objective

To estimate the direct medical costs and components of treating LN in the Brazilian private healthcare system

Methods

- A literature review was conducted to inform construction of a questionnaire to collect data to understand the disease characteristics, epidemiology, guideline recommendations and resources used for LN management
- A group of five specialists (composed of rheumatologists and nephrologists) participated in the project, which consisted of two main stages: filling out a preliminary questionnaire and a virtual meeting
- The selection and recruitment of specialists was carried out by a contracted company and was blinded both for the study sponsor (GSK) and for the participating specialists
- Initially, the specialists answered a questionnaire in order to identify the healthcare resources needed for diagnosis, prophylaxis (including parasitological examinations and tuberculosis screening), treatment, response and monitoring, kidney flares, dialysis, and kidney transplantation for patients with LN
- The use of resources was segmented according to the LN treatment profile being considered; specifically, classes III–IV and V, refractory LN (defined as those unresponsive to treatment), and ESKD
- Responses were obtained and the results were analyzed to guide discussion on the Expert Panel Day. The expert panel interview was conducted online, with a duration of 2 hours
- A final report was prepared containing information from the questionnaires and expert panel discussions
- For micro-costing, the average reported use of each resource and its respective unit cost were considered. Thus, the weighted total cost per patient was estimated over an annual time horizon, according to the respective LN class. Unit costs were obtained from Brazilian official price lists* (CBHPM 2020, D-TISS 2016 and CMED 2021; BRL 1 = US $5.64)
- It was assumed patients were in induction therapy for 3 months and, after reaching response in this period, they advanced to the maintenance period and would be managed accordingly in the remaining 9 months. This aligns with EULAR's 2019 recommendations, even though induction can last 6 months[6]

Results

- Data from the expert panel interview showed that approximately 76.0% of patients with LN were submitted to renal biopsy, of which 48.1% were in class III–IV LN and 21.4% in class V; 20.3% of patients had refractory LN and 10.3% had ESKD needing renal transplant
- Patients with LN classes III–IV had the highest frequency of renal flares (67.5%) with an annual mean of 4 episodes
- Average costs for LN diagnosis, prophylaxis, and response and monitoring were BRL 2564, BRL 1092, and BRL 3162, respectively, for all LN classes
- The total cost was estimated to be BRL 299,077 per patient with ESKD, representing the highest annual cost (**Figure 1**). The main cost driver was dialysis (BRL 220,434 [73.7% of the total]), followed by transplant (BRL 71,826 [24.0%]) (**Figure 2**)
- LN classes III–IV were the most prevalent and represented the second highest annual costs per patient, estimated to be BRL 110,734 (**Figure 1**). The main cost component was renal flares (BRL 56,166 [50.7% of the total]), followed by dialysis (BRL 28,668 [25.9%]) (**Figure 2**)

Figure 1. Estimated average annual cost per patient with LN

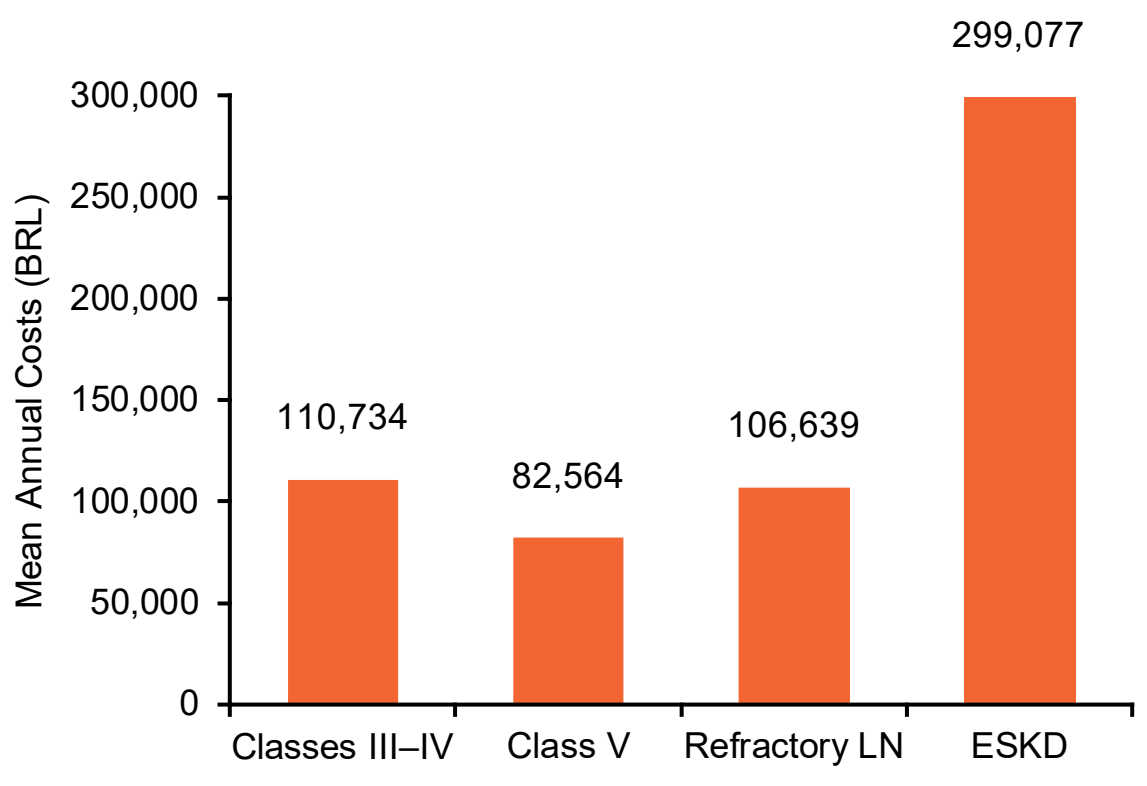
Mean Annual Costs (BRL)
300,000
250,000
200,000
150,000
100,000
50,000
0
110,734
82,564
106,639
299,077
Classes III–IV
Class V
Refractory LN
ESKD

Figure 2. Estimated average annual costs per patient per year by resource group

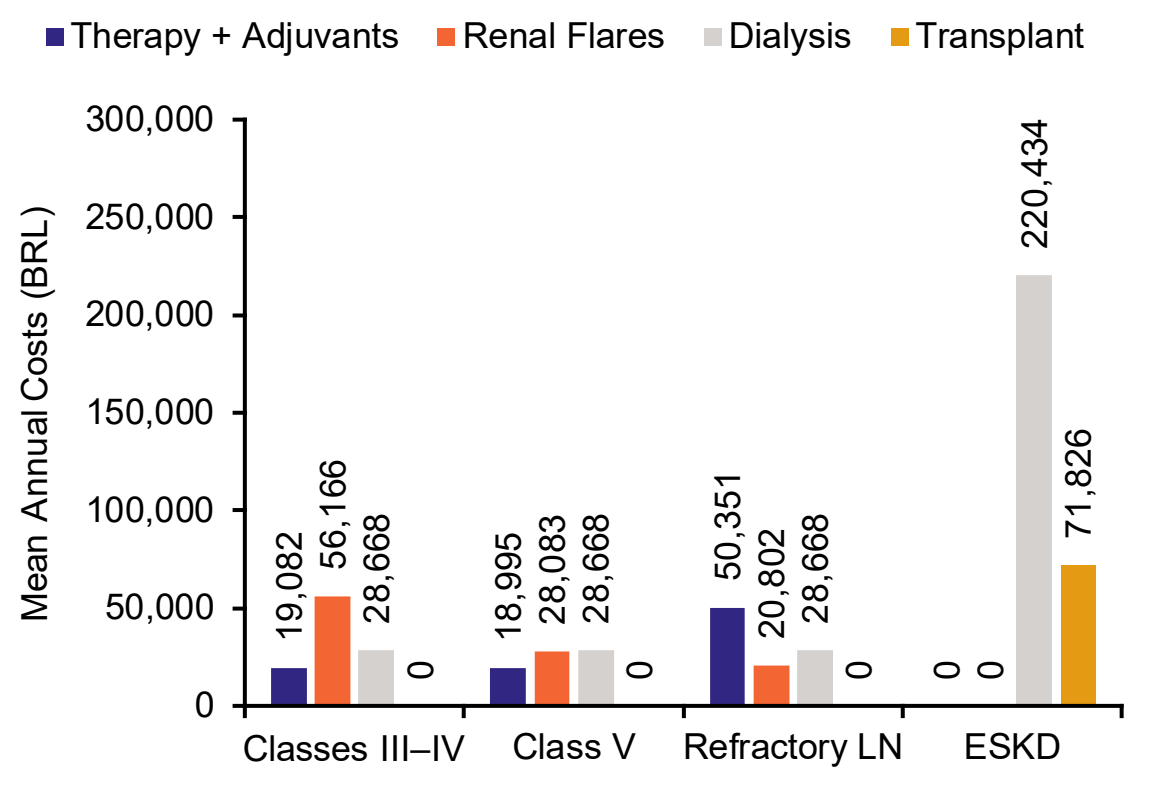
Therapy + Adjuvants
Renal Flares
Dialysis
Transplant
Mean Annual Costs (BRL)
300,000
250,000
200,000
150,000
100,000
50,000
0
19,082
56,166
28,668
0
18,995
28,083
28,668
0
50,351
20,802
28,668
0
0
0
220,434
71,826
Classes III–IV
Class V
Refractory LN
ESKD

- For patients with LN class III–IV, cyclophosphamide was the most used treatment for induction (66.0%), followed by mycophenolate mofetil (46.0%). For maintenance therapy, 50.0% of patients used mycophenolate mofetil and 48.0% used azathioprine
- Refractory LN was estimated to cost BRL 106,639 per patient; the main cost driver was therapy + adjuvants (BRL 50,351 [47.2%]) (**Figure 1 and 2**)
- Class V was estimated to cost BRL 82,564 per patient (**Figure 1**)

Conclusions

- The economic burden of LN management and treatment in the Brazilian private healthcare system is substantial for both the health system and patients
- With LN progression to most severe forms, particularly to ESKD, costs increase significantly

*Classificação Brasileira Hierarquizada de Procedimentos Médicos (CBHPM 2020) was used for medical appointments and procedures. Painel de Troca de Informações na Saúde Suplementar (D-TISS 2016) was used for hospitalization costs of complications. Câmara de Regulação do Mercado de Medicamentos (CMED 2021) was used for drug prices

Disclosures

DOS, **PS**, **ND**, **MRG**, and **GB** are employees of GSK. **ND**, **MRG** and **GB** own stocks/shares in GSK. **BK** and **DK** are employees of IQVIA and conducted the study on behalf of GSK. **ND** and **MRG** did not contribute to the development of this poster, but provided final approval

Acknowledgments

This study was funded by GSK (GSK study 217650). Editorial support for poster development was provided by Robert Bloxham, PhD, Fishawack Indicia Ltd, UK, part of Fishawack Health, and was funded by GSK.

References

1. Anders H-J, et al. *Nat Rev Dis Prim.* 2020;6(1):7.
2. Souza DCC, et al. *J Rheumatol.* 2012;39(3):496–503.
3. Murimi-Worstell IB, et al. *BMJ Open.* 2020;10(5):e031850.
4. Morales E, et al. *Nephron.* 2021;145(1):1–13.
5. Pelletier EM, et al. *Clin Ther.* 2009;31(11):2653–2664.
6. Fanouriakis A, et al. *Ann Rheum Dis.* 2020;79(6):713–723.

Author email address: danielle.o.silva@gsk.com

Presented at the ISPOR congress, May 15–18, 2022, online