Baseline Characteristics of Patients prior to Oriahnn Prescription

Fatema Turkistani[1], Beverly Johns[1] and Brett Pinsky[1]

[1]AbbVie Inc., North Chicago, IL, USA

OBJECTIVE

Describe the baseline demographics and clinical characteristics of the Oriahnn patient population.

CONCLUSIONS

Most patients using Oriahnn were over 40 years old, Caucasian, and have a lower annual income

Prior to starting Oriahnn, patients were most commonly treated with hormonal therapies

Lack of claims with a diagnosis of UF and/or HMB plus the presence of codes for a normal gynecological exam suggests that UF and HMB are not regularly coded in claims databases following initial diagnosis.

For additional information or to obtain a PDF of this poster

Scan QR code or utilize the following link to download an electronic version of this presentation and other AbbVie 2022 ISPOR scientific presentations:
https://abbvie1.outsystemsenterprise.com/GMAEventPublications/Assets.aspx?ConferenceId=340
QR code expiration: May 15, 2023
To submit a medical question, please visit
www.abbviemedinfo.com

AbbVie and the authors thank the participants, study sites, and investigators who participated in this clinical trial.

All authors had access to relevant data and participated in the drafting, review, and approval of this publication. AbbVie funded this trial and participated in the trial design, research, analysis, data collection, interpretation of data, and the review and approval of the publication. All authors had access to relevant data and participated in the drafting, review, and approval of this publication. No honoraria or payments were made for authorship.

Financial arrangements of the authors with companies whose products may be related to the present report are listed as declared by the authors: Brett Pinsky and Beverly Johns are full-time employees of Abbvie. Fatema Turkistani was a contracted employee at the time of the study.

International Society for Pharmacoeconomics and Outcomes Research (ISPOR); May 15 – 18, 2022

INTRODUCTION

- Uterine fibroids (also called leiomyomas or myomas) are non-cancerous, hormonally-responsive muscle tissue tumors of the uterus.[1]
- Fibroids can cause symptoms such as heavy menstrual bleeding (HMB), lower back and/or abdominal pain, pain during sex, increased frequency of urination, constipation, difficult bowel movements and abdominal cramps.[1,2]
- Treatment options for uterine fibroids include surgeries/procedures (e.g., hysterectomy, myomectomy, and endometrial ablation) and medical management with treatments (e.g., oral contraceptives, progestins, and gonadotropin-releasing hormone (GnRH) agonists).[1,3]

METHODS

- Descriptive retrospective study using Symphony Health (SHA) early view data
 - Medical and pharmacy claims data
- Inclusion Criteria
 - One prescription fill for Oriahnn from 07/01/2020 through 09/01/2021,
 - Age 18 and over
 - Continuously enrolled for 6 months prior to first Oriahnn fill.
- All Comorbidities and test were defined from the claims using standard codes.

RESULTS

Table 1. Baseline Characteristics of Oriahnn

Age; Mean (SD)	42.4 (6.8)
Age (Categorical)	N
18-29	79 (4.1%)
30-39	508 (26.5%)
40-49	1,067 (55.6%)
50-59	260 (13.6%)
60-64	3 (0.2%)
65+	1 (0.1%)
Race*	
Black	484 (38.4%)
Caucasian	560 (44.5%)
Other	215 (17.1%)
Income†	
\$ 0 - \$ 29,999	407 (33.1%)
\$ 30,000 - \$ 49,999	210 (17.1%)
\$ 50,000 - \$ 74,999	194 (15.8%)
\$ 75,000 - \$ 99,999	141 (11.5%)
\$100,000 +	278 (22.6%)

*659 Undefined Race ꟷ †688 Undefined Income

Baseline Characteristics

- A total of 1,918 patients have been enrolled in the study
- Average age of patients was 42.4
 - Over half were between the 40-49
- Race and Income information was available for ~65% of the populations
 - 38% of the patients were Black
 - 1/3 of the patients were low income
- The number of comorbid conditions among Oriahnn patients was low; but they still on average had 2 outpatient visits in the 6 months prior to starting Oriahnn (Table 2)

Table 2. Utilization prior to Oriahnn Start

	Mean (SD)
Baseline Charlson Comorbidity Score	0.1 (0.4)
Baseline Health Care visits	
Outpatient	2.3 (3.9)
Inpatient	0 (0.2)
Baseline Medication Costs Per Month	\$55.7 (\$243.7)

Table 3. Most Common Diagnosis, Prescription and Procedures in the Baseline

Diagnosis		Drug Class		Procedure	
Leiomyoma of uterus, unspecified	359 (18.7%)	Sex hormones	534 (27.8%)	Ultrasound	305 (15.9%)
Excessive and frequent menstruation with regular cycle	262 (13.7%)	Miscellaneous coagulation modifiers	188 (9.8%)	Blood Draw	281 (14.7%)
Essential (primary) hypertension	185 (9.6%)	Antineoplastic hormones	127 (6.6%)	Biopsy	143 (7.5%)
Encounter for gynecological examination without abnormal findings	169 (8.8%)				
Anemia, unspecified	146 (7.6%)				

- Uterine Fibroid (Leiomyoma of uterus) and Excessive menstruation (Heavy Menstrual Bleeding) were the most common diagnoses and nearly 9% of Oriahnn patients had a claim indicating a normal gynecological examination
- The most common treatments in the baseline period were hormone therapy, and the most common procedures were diagnostic procedures
- Oriahnn use is increases over time (Figure 1)

Figure 1. Distribution of Index Date Quarter and Year

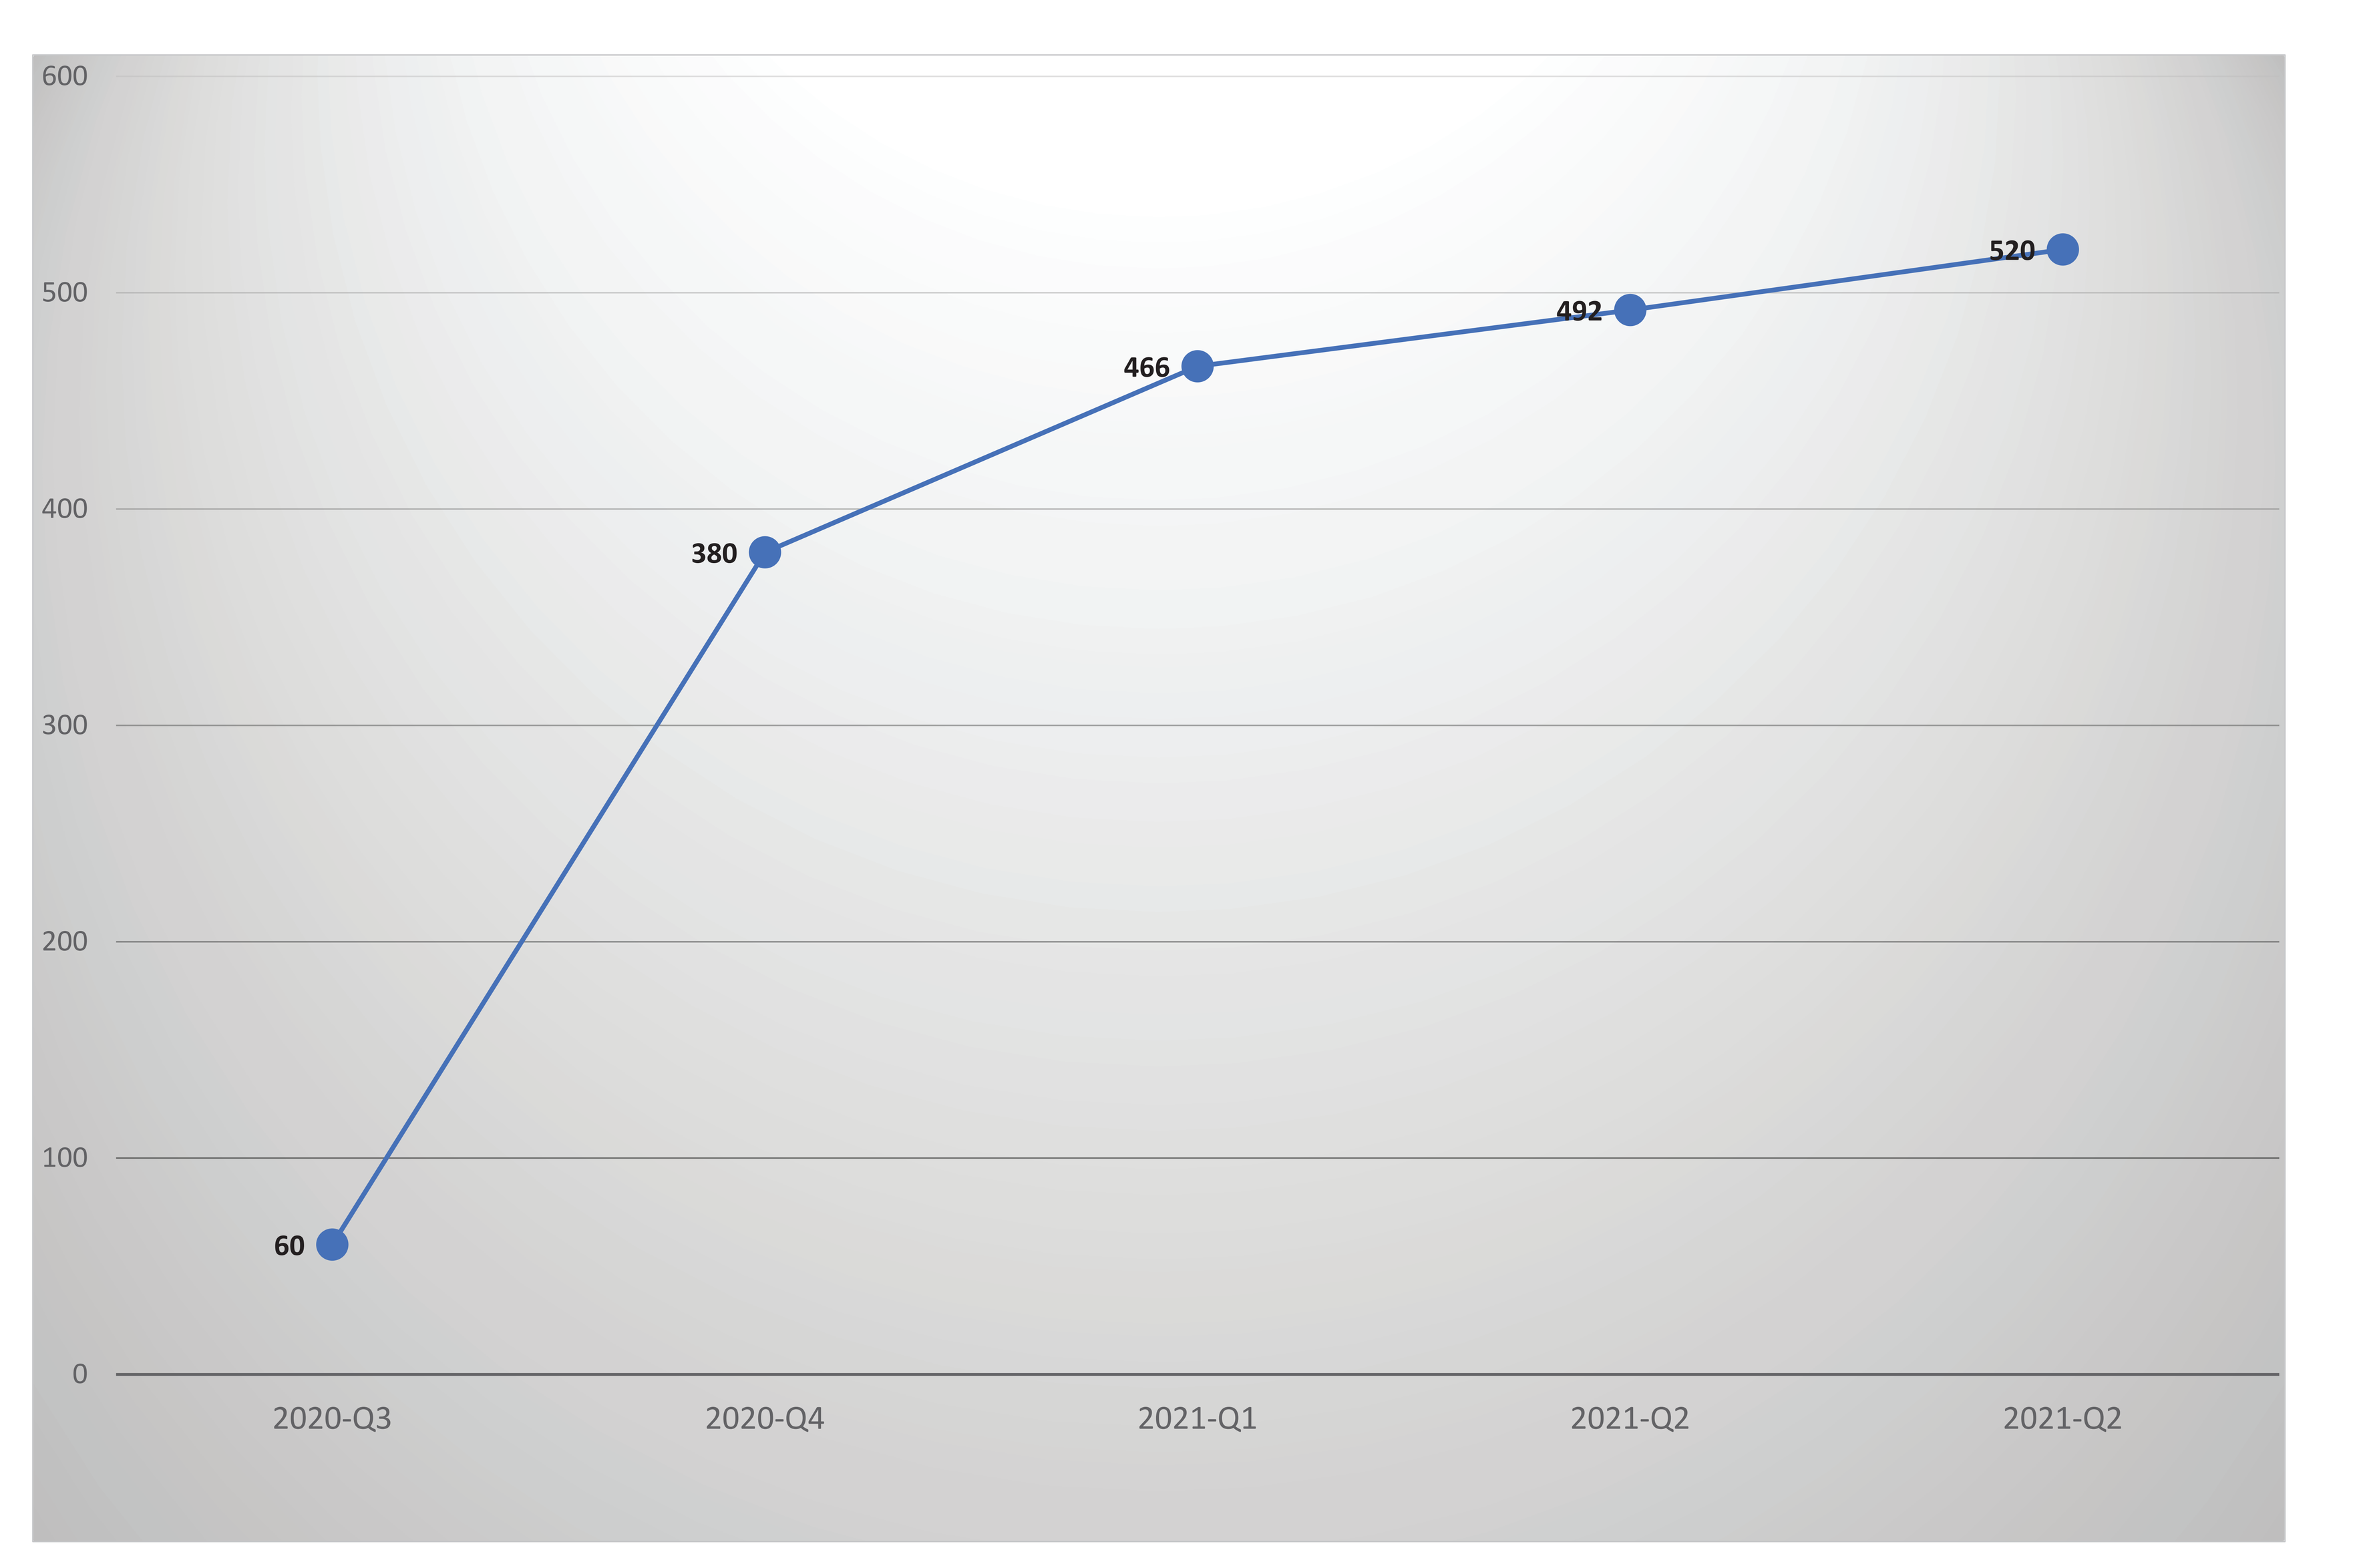
600
500
400
300
200
100
0
60
380
466
492
520
2020-Q3
2020-Q4
2021-Q1
2021-Q2
2021-Q2

1. De La Cruz MS, Buchanan EM. Uterine Fibroids: Diagnosis and Treatment. Am Fam Physician. 2017 Jan 15;95(2):100-107.
2 The American College of Obstetricians and Gynecologists: FAQ Uterine Fibroids. https://www.acog.org/patient-resources/faqs/gynecologic-problems/uterine-fibroids. Accessed March 23, 2020.
3 Alternatives to hysterectomy in the management of leiomyomas. ACOG Practice Bulletin No. 96. American College of Obstetricians and Gynecologist. Obstet Gynecol 2008; 112:387-400.