Health disparities in health technology assessment: An opportunity for improvement

Ben Penley, PharmD[1]; Jane Ha, PharmD[1]; Evelyn Sarnes, PharmD, MPH[1]
[1]Xcenda LLC, Carrollton, TX

Background

- Traditionally, health technology assessment (HTA) bodies have evaluated technologies based on efficiency to determine their value and inform resource allocation.
- Equity in the provision of healthcare and access to health resources is vital, and payers and HTAs are in a position to support equitable access to technology within their member populations.
- The World Health Organization (WHO) defines **health equity** as "the absence of unfair, avoidable, and remediable differences in health status among groups of people" that is achieved "when everyone can attain their full potential for health and well-being."[1]
- **Health disparities** are defined as "a particular type of health difference that is closely linked with social, economic, and/or environmental disadvantage," adversely impacting certain groups of people who have "systematically experienced greater social or economic obstacles to health" based on race, socioeconomic status, or other characteristics associated with discrimination or exclusion.[2]
- Health equity can be seen as the overarching "social justice in health," while health disparities are the "metric [used] to measure progress toward achieving health equity."[3] Ultimately, striving toward equitable access includes minimizing health disparities in order to achieve greater health equity.

Objective

- To determine if and how major HTA bodies consider health disparities in their topic selection and assessment of new technologies.

Methods

- HTA bodies in North America, Europe, and Australia were identified that have formal submission requirements and make public their appraisals:
 - National Institute for Health and Care Excellence (NICE) (United Kingdom [UK])
 - Scottish Medicines Consortium (SMC) (Scotland)
 - Institute for Quality and Efficiency in Health Care (IQWiG; *Institut für Qualität und Wirtschaftlichkeit im Gesundheitswesen*) (Germany)
 - Pharmaceutical Benefits Advisory Committee (PBAC) (Australia)
 - Canadian Agency for Drugs and Technologies in Health (CADTH) (Canada)
 - Institute for Clinical and Economic Review (ICER) (US)
 - Academy of Managed Care Pharmacy (AMCP) (United States [US]) (Note: AMCP dossiers are not available publicly)
- For each HTA body, we evaluated their most recent guidance documents that detailed their assessment processes, formal submission requirements for manufacturers, and parameters for topic selection. Documents were identified on each HTA's website and screened independently by 2 researchers to determine if and how health disparities were discussed.
- We then identified health conditions in which there are recognized health disparities and recently approved treatments. We reviewed publicly available appraisals for these technologies: crizanlizumab (sickle cell anemia), alirocumab (hypercholesterolemia), and buprenorphine extended-release injections (opioid use disorder).

Results

HTA submission requirements and value frameworks

- 6 of the 7 HTA bodies reviewed considered equity to an extent in their guidance (**Table 1**).
- Of the 7 HTA bodies reviewed, 3 (CADTH, AMCP, IQWiG) did not request information related to disparities as part of the submission, while 3 (NICE, SMC, PBAC) did encourage manufacturers to discuss their technology in relation to existing disparities with an intent to reduce health disparities (**Table 1**).
- No HTA bodies formally integrated equity considerations in comparative effectiveness analyses.
- 6 HTA bodies solicited patient input at early stages in their assessments, during which discussion of health disparities might arise.

Table 1. HTA bodies' equity considerations and manufacturer submission requirements

HTA	Equity considerations in guidance	Requests manufacturer to include equity considerations in submission?
AMCP	• No explicit considerations	No
IQWiG	• Subgroup analyses for gender and age	No
CADTH	• Subgroup analyses defined by equity-related characteristics • Identification of groups that are likely to be disadvantaged by the adoption or implementation of a technology • Discussion of equity-efficiency trade-offs	No
NICE	• Subgroup analyses considering equality • NICE equality scheme to eliminate unlawful discrimination and promoting equality of opportunity • In economic evaluations, all QALYS are considered of equal value in the reference case • "Equality considerations" section in template	Yes
SMC	• Notes difficulty of including equity considerations in economic evaluation; all QALYs are considered of equal value in the reference case	Yes
PBAC	• Notes that equity should influence PBAC decision making • Excludes changes in production as an outcome of therapy in base-case economic analyses due to equity implications	Yes
ICER	• Scenario analyses to capture a technology's impact on disparities in life expectancy across subpopulations • Topics for review are selected in consideration of equity • Notes that equity considerations are challenging to quantify • "Society's goal of reducing health inequities" section in template, on which members vote to determine the technology's impact on this domain	Does not request manufacturer submissions

Review of specific product HTA appraisals

- In the appraisals of crizanlizumab, alirocumab, and buprenorphine, information on health disparities was included in 9 of 15 HTA reports.
- Most of the appraisals recognized potentially disadvantaged populations in the respective diseases, acknowledged equal access to care concerns, or identified gaps in representation in clinical studies (**Table 2**).
 - NICE and ICER most consistently considered health disparities.
 - In all products reviewed, NICE included an "equality considerations" section.
 - In all products reviewed, ICER included, and members voted on, the importance of the intervention for the criteria, in a "potential other benefits – society's goal of reducing health inequities" section.
- No HTA bodies formally integrated or weighted health disparities in determining the cost-effectiveness of the treatment or in the final recommendation. However, for crizanlizumab, both NICE and ICER discussed that higher cost-effectiveness ratios may be warranted if the product could reduce health inequalities (**Table 2**).
 - Ultimately, NICE did not recommend crizanlizumab because of uncertainty about the product's long-term clinical effectiveness.
- It is important to note that the discussion of health disparities in the appraisals may have come from the manufacturer's own submissions, may have been part of the HTA body's consideration of the product, or could have been raised during patient input.

Table 2. Discussion of equity in publicly available HTA reports

	Crizanlizumab	Alirocumab	Subcutaneous buprenorphine
AMCP[a]	-	Y	-
IQWiG	N	N	-
CADTH	-	N	N
NICE	Y	Y	Y
SMC	-	N	Y
PBAC	-	Y	N
ICER	Y	Y	Y

- **NICE on crizanlizumab:** *The committee said that* ***in theory it would be willing to accept an [incremental cost-effectiveness ratio] slightly more than what is usually acceptable if it addresses such health inequalities.*** *However, it noted that departing from NICE's usual range needs to be done with caution, as it risks displacing funding from more cost-effective treatments elsewhere in the [National Health Service], with an overall net loss of health gain.*
- **ICER on crizanlizumab:** *ICER notes that decision makers in the US may wish to* ***consider giving special weighting to other benefits and to contextual considerations [such as improving equitable access to care] that would lead to coverage and funding decisions at higher prices, and thus higher [cost-effectiveness] ratios****, than applied to other decisions about other treatments.*
- **PBAC on alirocumab:** *The population with [cardiovascular disease] is not homogenous. Factors identified [by the manufacturer] as being important to the safe and effective use of alirocumab did not address the* ***different considerations for populations with the highest burden of [cardiovascular disease], including Aboriginal and Torres Strait Islanders and those with mental illness.***
- **SMC on SC buprenorphine:** *Clinical experts considered that the place in therapy could be in* ***patients who have difficulty attending daily pharmacy visits; for example, due to mobility problems, those working in education, family commitments, or irregular attendance****. The reduced frequency of administration could support social integration.*

[a]AMCP dossiers were not publicly available. However, the alirocumab dossier was made available through a relationship with the manufacturer.

Discussion

Equity and efficiency concepts in HTA

- "Equity checklists" have been proposed that include important considerations and questions to inform HTAs—from scoping to final recommendations.
- However, before integrating health equity considerations into the HTA process, the importance and appropriate balance of equity in relation to efficiency should be considered. Modern HTA is focused on efficiency—that is, how to maximize population health within finite resources (economical distribution)—whereas the concept of equity advocates for the fair and unbiased distribution of healthcare.
 - HTA bodies must grapple with not only considering whether a technology will improve equitable access for a known disparity (eg, accepting a higher cost-effectiveness ratio), but what the impact of the higher-cost treatment will be on the allocation of remaining resources.
- The literature contains numerous conceptual methods to formally integrate health equity considerations into HTA decision making:
 - Multi-criteria decision analysis (MCDA) is one proposed method to integrate health equity considerations in HTAs in which a group of stakeholders identify and weigh criteria—qualitatively or quantitatively—like "equity" and "efficiency" according to their importance.[6-8]
 - Health equity can also be formally integrated into cost-effectiveness analyses by weighting QALYs based on equity-related characteristics of a group, expressing the extent to which society is willing to trade health benefits for a more equitable distribution of health.[9]

The future of health disparities in HTA

- The transition to a European Joint Clinical Assessment (JCA) was initiated in January 2022, with the aim to create a centralized process for technology assessments across Europe.[10]
 - The Core Model of the European Network for Health Technology Assessment (EUnetHTA) provides a framework to facilitate international collaboration in HTAs.
 - The "ethical analysis" and "patients and social aspects" domains in the core model reflect on disparities in their respective assessment elements, "justice and equity" and "social group aspects," posing the questions:
 - How does implementation or withdrawal of the technology affect the distribution of healthcare resources?
 - How are technologies with similar ethical issues treated in the healthcare system?
 - Are there factors that could prevent a group or person from gaining access to the technology?
 - Are there groups of patients who currently don't have access to available therapies?
- With the recent legal framework for the JCA, health disparities may become a core component of submissions and appraisals.

Conclusion

- Though no HTA bodies have formally integrated equity considerations in their comparative effectiveness analyses, several have expressed their importance, requested manufacturers include considerations for disparities in their submissions, and considered them in their technology assessments.
- Most HTA bodies do seek patient input, which is an important consideration in the HTA process. Patient input can have implications in how HTA bodies consider health equity in their appraisals, as patient groups might raise awareness of these concerns. However, relying on patient input to address equity in HTAs alone is not sufficient, as it places the onus on patient groups and can lead to an ad hoc approach to equity considerations.
- In the position to support equitable access to technology within their member populations, HTA bodies should work to—at the least—qualitatively consider equity in their assessments.

References

1. World Health Organization. Health equity and its determinants. 2021. Accessed March 31, 2022. https://cdn.who.int/media/docs/default-source/world-health-day-2021/health-equity-and-its-determinants.pdf
2. Office of Disease Prevention and Health Promotion. Disparities. February 6, 2022. Accessed March 31, 2022. https://www.healthypeople.gov/2020/about/foundation-health-measures/Disparities
3. Braveman P. What are health disparities and health equity? We need to be clear. *Public Health Rep*. 2014;129 (Suppl 2):5-8.
4. Cuyler AJ, Bombard Y. An equity framework for health technology assessments. *Med Decis Making*. 2012;32(3):428-441.
5. Benkhalti M, Espinoza M, Cookson R, Welch V, Tugwell P, Dagenais P. Development of a checklist to guide equity considerations in health technology assessment. *Int J Technol Assess Health Care*. 2021;37(e17):1-8.
6. Johri M, Norheim OF. Can cost-effectiveness analysis integrate concerns for equity? Systematic review. *Int J Technol Assess Health Care*. 2012;28(2):125-132.
7. Baltussen R, Marsh K, Thokala P, et al. Multicriteria decision analysis to support health technology agencies: benefits, limitations, and the way forward. *Value Health*. 2019;22(11):1283-1288.
8. Marsh KD, Sculpher M, Caro JJ, Tervonen T. The use of MCDA in HTA: great potential, but more effort needed. *Value Health*. 2018;23(4):394-397.
9. Bobinac A, van Exel NJA, Rutten FFH, Brouwer WBF. Inquiry into the relationship between equity weights and the value of the QALY. *Value Health*. 2012;15:1119-1126.
10. EUnetHTA. HTA Core Model Version 3.0. January 25, 2016. Accessed March 31, 2022. http://www.corehta.info/model/HTACoreModel3.0.pdf

Acknowledgments: The authors would like to thank Seth Cook, Robin Tan, Thao Luu, Marie-Josee Martel, Werner Kulp, and Ken O'Day for their contributions and input to this project.

Presented at ISPOR 2022
May 15-18, 2022 | Washington, DC
Funded by Xcenda

AmerisourceBergen
Xcenda