Treatment patterns, clinical outcomes, and healthcare resource use among patients with stage IV urothelial cancer: findings from a nationwide cohort study in Denmark

S. Sørup,[1] B. Darvalics,[1] M. Kearney,[2] S.H. Mahmoudpour,[2] D. Oksen,[2] F. Kolitosopoulos,[3] M. Agerbaek,[4] D. Cronin-Fenton[1]

[1]Aarhus University, Aarhus, Denmark; [2]Merck KGaA, Darmstadt, Germany; [3]Pfizer, New York, NY, USA; [4]Aarhus University Hospital, Aarhus, Denmark

SCOPE

- **To characterize treatment patterns, survival outcomes, and healthcare resource use in patients with stage IV urothelial carcinoma (UC) in Denmark**

CONCLUSIONS

- **This observational cohort study describes the characteristics of an older and relatively frail population with stage IV UC in a real-world setting in Denmark**
- **A considerable proportion of patients did not receive any systemic anticancer treatment including PD-L1/PD-1 immune checkpoint inhibitors, reflecting the limited availability of these agents in standard care during the study period**
- **Patients with stage IV UC had substantial healthcare resource use across all sites of care investigated in this study**
- **There is a large unmet medical need for new therapeutic options for elderly patients with significant comorbidities and poor outcomes**
- **This study can serve as a useful benchmark for future real-world studies in stage IV UC, thereby enabling clinicians to better understand the utilization of and outcomes associated with newer systemic agents**

GET POSTER PDF

Copies of this poster obtained through this **hyperlink** or quick response (QR) code are for personal use only and may not be reproduced without permission from ISPOR 2021 and the author of this poster.

Correspondence: Mairead Kearney, **mairead.kearney@merckgroup.com**

BACKGROUND

- Bladder cancer is the 10th most common cancer globally; in 2018, >549,000 new cases were reported and nearly 200,000 patients died from the disease worldwide[1]
- Nearly 204,000 people in Europe were diagnosed with bladder cancer across all stages in 2020, and more than 67,000 patients died from the disease, despite available treatments[2]
- UC accounts for approximately 90% of all bladder cancers[3]
- Approximately 5% of patients with UC are diagnosed with distant metastases; in these patients, the 5-year overall survival (OS) rate is <6%[4]
- Platinum-based combination chemotherapy (PBC) remains the standard first-line (1L) treatment for patients with advanced UC, ie, stage III/IV disease[5]
 - Although response rates to PBC regimens are high (70%), durable response is limited, and most patients ultimately experience disease progression
- The goal of treatment for patients with advanced UC is to prevent disease progression, maintain health-related quality of life (HRQOL), provide relief from cancer symptoms, and extend life

METHODS

Study design

- This cohort study used information from Danish population-based and medical registries
- Patients diagnosed with incident stage IV UC and registered in the Danish Cancer Registry[6] from January 1, 2013, to December 31, 2017, were included and followed-up until December 31, 2018

Data sources

- OS was calculated using the date of all-cause death from the Danish Civil Registration System[7]
- Information about tests for PD-L1 biomarkers was obtained from the Danish National Pathology Registry[8]
- Information about anticancer treatment and health resource utilization was obtained from the Danish National Patient Registry[9]

Ethics

- Under Danish law, ethics committee permission is not required for registry-based research. This study followed the EU General Data Protection Regulation. Data was obtained from the Danish Health Data Authority

Statistical analyses

- Summary and descriptive statistics were calculated for categorical and continuous variables as appropriate
- The Kaplan-Meier method was used to estimate median OS
- SAS version 9.4 was used for analyses, and R version 4.0.2 was used for visualizations

RESULTS

Figure 1. Patient attrition

Bladder and UC diagnosed in 2013-2017 according to the DCR (ICD-10 codes: C65-C68.0) and included in the DCRS at the time of diagnosis
n=5,369

No AJCC stage II, III, or IV at the time of diagnosis
n=2,488

AJCC stage II, III, or IV at the time of diagnosis
n=2,881

No morphology codes* for first-time† transitional cell carcinoma
n=377

No AJCC stage IV at the time of diagnosis
n=1,884

Included patients with **UC** AJCC stage IV at the time of diagnosis
n=620

The population for this study was a subpopulation of an ongoing study including 2,504 patients with UC with at least AJCC stage II at the time of diagnosis.
AJCC, American Joint Committee on Cancer; **DCR**, Danish Cancer Registry; **DCRS**, Danish Civil Registration System; **UC**, urothelial cancer.
*ICD-O3 morphology codes different from 81203, 81223, 81313.
†Sixteen patients had 2 registrations with bladder or urothelial transitional cell carcinoma AJCC stage II, III, or IV in the DCR; we excluded the latest of these registrations.

Patients

- A total of 620 patients met inclusion criteria and were included in this analysis
 - Patient characteristics are shown in **Table 1**
- The median age was 72.2 years; 474 patients (76.5%) were aged ≥65 years, 394 (63.5%) were men, and 410 (66.1%) had a baseline Deyo-Charlson Comorbidity Index score of ≤2
- The median follow-up was 10.2 months (IQR, 3.6-19.4 months)
- PD-L1 expression testing was not routinely conducted; only 9 patients (1.5%) were tested

Table 1. Patient characteristics

Patients with incident stage IV UC (stage IV)	N=620
Age at diagnosis, years	
Mean (SD) Median (Q1-Q3)	71.6 (9.6) 72.2 (65.6-78.2)
Age category, n (%)	
35-49 years 50-64 years ≥65 years	15 (2.4) 131 (21.1) 474 (76.5)
Sex, n (%)	
Male Female	394 (63.5) 226 (36.5)
Deyo-Charlson Comorbidity Index, n (%)[10,*]	
0 1-2 ≥3	308 (49.7) 102 (16.5) 210 (33.9)
Duration of follow-up from diagnosis, months	
Mean (SD) Median (Q1-Q3)	14.4 (14.8) 10.2 (3.6-19.4)
Biomarkers†	
PD-L1 expression, n (%) Not tested Tested	 611 (98.5) 9 (1.5)

*We included information on diagnoses from 1977 (start date of the Danish National Patient Register) until and including date of meeting inclusion criteria for the specified cohort (date of cancer diagnosis). We excluded diagnoses of cancer when calculating the Deyo-Charlson Comorbidity Index.
†Biomarkers were examined from 90 days before the date of cancer diagnosis until the end of follow-up because some tests might be performed as part of the diagnostic procedures.

Treatment patterns

- 134 patients (21.6%) received no anticancer treatment (ie, no therapeutic surgery, radiation therapy, or medical therapy)
 - 353 patients (56.9%) underwent ≥1 therapeutic surgical procedure; 193 (31.1%) received radiation therapy

Systemic anticancer therapy

- 299 patients (48.2%) received systemic anticancer therapy during follow-up
- The most commonly administered drug classes in 1L systemic treatment were platinum-containing agents (69.6%) and antimetabolites (83.6%)
- Only a minority of patients (n=30 [4.8%]) received ≥1 administration of PD-L1/PD-1 immune checkpoint inhibitors

Healthcare resource utilization

- The mean (SD) per-patient per-year number of outpatient visits, hospital admissions, and emergency department visits was 26.6 (24.4), 7.6 (7.3), and 3 (3.5), respectively (**Table 2**)

Table 2. Healthcare resource utilization

Urothelial carcinoma, AJCC stage IV, healthcare utilization from index date	N=620
Healthcare utilization, PPPY	
PPPY outpatient visits	
Mean (SD) Median (Q1-Q3)	26.6 (24.4) 20.9 (10.4-35.0)
PPPY hospitalizations	
Mean (SD) Median (Q1-Q3)	7.6 (7.3) 5.5 (2.8-10.3)
PPPY emergency department visits	
Mean (SD) Median (Q1-Q3)	3 (3.5) 1.6 (0.84-3.7)

AJCC, American Joint Commission on Cancer; **PPPY**, per patient per year; **Q1**, first quartile; **Q3**, third quartile.

Overall survival

- Median OS from diagnosis was 9.4 months (95% CI, 8.3-10.6 months) (**Figure 2**)
 - The 12- and 24-month survival rates were 40.4% (95% CI, 36.4%-44.4%) and 23.6% (95% CI, 20.1%-27.4%), respectively

Figure 2. OS from the date of diagnosis of stage IV UC (N=620)

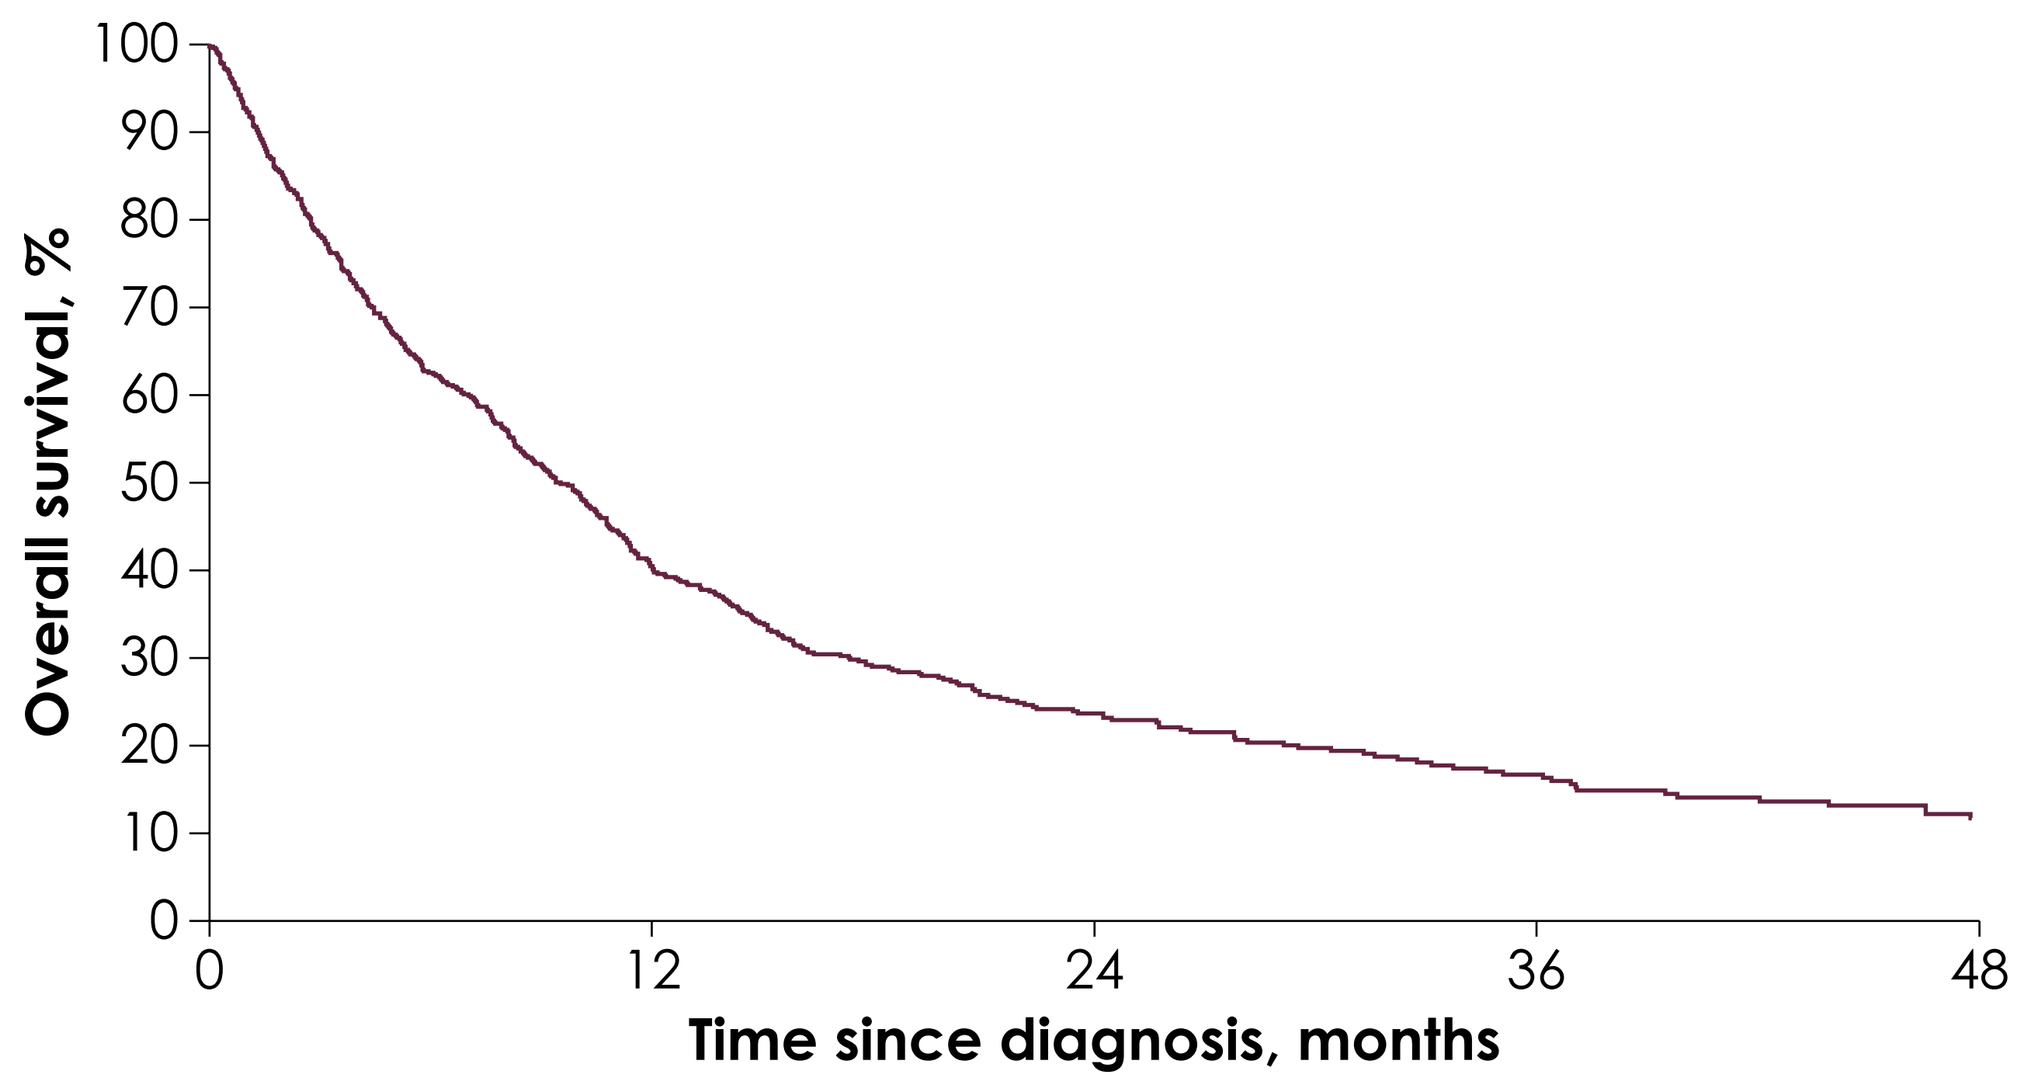
Overall survival, %
100
90
80
70
60
50
40
30
20
10
0
0
12
24
36
48
Time since diagnosis, months

Strengths and limitations

- Strengths
 - This study includes nationwide registries, representing real-world clinical practice in Denmark
 - Data represent the full continuum of care with documented and longitudinal tracking of patient information
- Limitations
 - Limited information was available on disease-specific outcomes, such as cancer recurrence and disease progression
 - No information was available on number of lines of systemic therapy

REFERENCES 1. Bray F, et al. CA Cancer J Clin. 2018;68(6):394-424. **2.** IARC. Bladder fact sheet: GLOBOCAN. Accessed March 15, 2021. https://gco.iarc.fr/today/data/factsheets/cancers/30-Bladder-fact-sheet.pdf. **3.** Cancer.net. Bladder cancer: introduction. Accessed March 2021. https://www.cancer.net/cancer-types/bladder-cancer/introduction. **4.** Surveillance, Epidemiology, and End Results Program. Cancer Stat Facts: bladder cancer. https://seer.cancer.gov/statfacts/html/urinb.html. Accessed 15, March 2021. **5.** National Comprehensive Cancer Network. NCCN Clinical Practice Guidelines in Oncology: bladder cancer. Version 1.2021. Accessed 15, March 2021. **6.** Gjerstorff ML. Scand J Public Health 2011;39(7 suppl):42-5. **7.** Schmidt M, et al. Eur J Epidemiol. 2014;29(8):541-9. **8.** Erichsen R, et al. Clin Epidemiol. 2010;2:51-6. **9.** Schmidt M, et al. Clin Epidemiol. 2015;7:449-90. **10.** Charlson ME, et al. J Chronic Dis. 1987;40(5):373-83. **DISCLOSURES M. Kearney**, **S.H. Mahmoudpour**, and **D. Oksen** are employees of Merck KGaA, Darmstadt, Germany. **F. Kolitosopoulos** is an employee of Pfizer. **M. Agerbaek** is an employee of Aarhus University Hospital, Aarhus, Denmark. **S. Sørup**, **B. Dervalics**, and **D. Cronin-Fenton** are employees of Aarhus University, Aarhus, Denmark, which received funding from Merck KGaA and Pfizer. **ACKNOWLEDGMENTS** This work was partly supported by Merck KGaA, Darmstadt, Germany, by an institutional grant to and administered by Aarhus University Hospital, as part of an alliance between Merck KGaA, Darmstadt, Germany, and Pfizer. Editorial support was provided by ClinicalThinking and was funded by Merck KGaA and Pfizer.

Poster No. PCN242. Presented at Virtual ISPOR 2021, May 17-20, 2021.