Economic Burden of Schizophrenia among Young Adults with Medicaid Coverage

Marie-Hélène Lafeuille,[1] Charmi Patel,[2] Dominic Pilon,[1] Maryia Zhdanava,[1] Dee Lin,[2] Aurélie Côté-Sergent,[1] Carmine Rossi,[1] Patrick Lefebvre,[1] Kruti Joshi[2]

[1]Analysis Group, Inc., Montréal, QC, Canada; [2]Janssen Scientific Affairs, LLC, Titusville, NJ, USA

BACKGROUND

- Schizophrenia is a chronic mental illness with onset typically occurring in young adulthood.[1] It is estimated to affect about 1% of adults in the United States (US)[2]
- The economic burden to the healthcare system and payers associated with schizophrenia in the US was estimated in a 2013 analysis to exceed $155.7 billion annually[3]
- In a commercially insured population, young adults (18-35 years of age) with schizophrenia demonstrated greater inpatient medical costs compared with older adults with schizophrenia[4]
- Compared with the overall US population, Medicaid beneficiaries are disproportionately affected by schizophrenia, with an estimated 2.9% suffering from the condition
- Recent data on the economic burden of schizophrenia in young adults covered by Medicaid are scarce

OBJECTIVES

- To evaluate incremental healthcare resource utilization (HRU) and costs among young adults with Medicaid coverage and schizophrenia relative to schizophrenia-free controls

METHODS

Data Source

- A 6-state Medicaid database including data from Iowa (Q1 1998 to Q3 2016), Kansas (Q1 2001 to Q1 2018), Mississippi (Q1 2006 to Q1 2018), Missouri (Q1 1997 to Q1 2018), New Jersey (Q1 1997 to Q1 2014), and Wisconsin (Q1 2004 to Q4 2013) was used
- Information on Medicaid enrollment and demographic variables, as well as claims for medical visits and prescription drugs, including corresponding costs, was available

Study Design

- A retrospective matched cohort design was used
 - Patients with schizophrenia were matched to patients without schizophrenia
 - Afterwards, the analysis was restricted to patients with schizophrenia aged 18-35 years old and their matched controls without schizophrenia
- The index date was the last observed diagnosis of schizophrenia (cohort with schizophrenia) or the last service claim (control cohort) with ≥12 months of continuous Medicaid enrollment before and after it
- Baseline characteristics were described in the 12-month pre-index (ie, baseline) period; HRU and costs were described and compared during the 12-month post-index (ie, follow-up) period

Inclusion Criteria

- Cohort with schizophrenia
 - ≥2 claims on different days with a diagnosis of schizophrenia (International Classification of Diseases, 9th Revision, Clinical Modification [ICD-9 CM] code: 295.xx; International Classification of Diseases, 10th Revision, Clinical Modification [ICD-10-CM] code: F20.xx, F21, F25.xx) during the last continuous Medicaid enrollment period
 - ≥1 claim with a diagnosis of schizophrenia with ≥12 months of continuous Medicaid enrollment before and after the claim, with the latest claim being the index date
- Control cohort
 - No claims with a diagnosis of schizophrenia during the last continuous Medicaid enrollment period
 - ≥1 service claim with ≥12 months of continuous Medicaid enrollment before and after the claim, with the latest claim being the index date
- Both cohorts
 - Aged ≥18 years as of the index date; young adults (18-35 years old) with schizophrenia and their matched controls without schizophrenia were subsetted from this population after matching

Figure 1. Identification of the study population[a]

Cohort with schizophrenia

≥2 claims with a diagnosis of schizophrenia during the last continuous Medicaid enrollment period (ICD-9-CM: 295.xx; ICD-10-CM: F20.xx, F21, and F25.xx)
N = 312,495

↓

≥1 claim with a diagnosis of schizophrenia with ≥12 months of continuous Medicaid enrollment before and after the claim, with the latest claim being the index date
N = 162,218 (51.9%)

↓

Patients with schizophrenia

Aged ≥18 years as of the index date
N = 158,763 (97.9%)

↓

Young adults with schizophrenia

Aged 18 to 35 years as of the index date
N = 29,271 (18.4%)

Control cohort without schizophrenia

Randomly selected patients without claims with a diagnosis of schizophrenia during the last continuous Medicaid enrollment period
N = 6,000,000

↓

≥1 claim with ≥12 months of continuous Medicaid enrollment before and after the claim, with the latest claim being the index date
N = 2,333,889 (38.9%)

↓

Patients without schizophrenia

Aged ≥18 years as of the index date
N = 1,166,758 (50.0%)

AP, antipsychotic; ICD-CM-9, International Classification of Diseases, 9th Revision, Clinical Modification; ICD-10-CM, International Classification of Diseases, 10th Revision, Clinical Modification.
[a]Patients with schizophrenia were matched to patients without schizophrenia. The analysis was then restricted to patients with schizophrenia aged 18-35 years and their matched controls without schizophrenia.

Outcome Measures

- HRU and healthcare costs were reported per-patient-per-year over the 12-month follow-up period based on medical and prescription drug claims
- Costs were adjusted for inflation using the medical care component of the US Consumer Price Index and were reported in 2018 US dollars

Statistical Analysis

- Patients with and without schizophrenia were matched 1:1 using exact matching factors (ie, age group) and propensity scores generated based on sex, race, state, insurance eligibility (ie, capitated versus dual Medicaid/Medicare coverage), and year of the index date
- The analysis was then restricted to patients with schizophrenia aged 18-35 years as of the index date and their matched schizophrenia-free controls
- HRU was compared between matched pairs of young adults with and without schizophrenia using Poisson regression models, and nonparametric bootstrap procedures were used to generate 95% confidence intervals (CIs) and *P* values
- Mean cost differences between matched pairs were estimated using ordinary least squares regression models, and nonparametric bootstrap procedures were used to generate 95% CIs and *P* values
- The comparisons of HRU and costs were adjusted for characteristics that remained unbalanced between the matched pairs (ie, race, state, and insurance eligibility)

RESULTS

Study Sample

- Among 158,763 patients with schizophrenia aged 18 years and older, 29,271 (18.4%) were young adults between 18 and 35 years of age (**Figure 1**); their characteristics are presented in **Table 1**

Table 1. Baseline characteristics in young adults with schizophrenia and their matched pairs without schizophrenia[a]

	Young Adults With Schizophrenia N = 29,271	Young Adults Without Schizophrenia N = 29,271	Standardized Difference (%)
Age at index date, mean ± SD (median), years	28.0 ± 4.6 (28.6)	27.5 ± 5.0 (28.2)	11.6
Female, n (%)	11,160 (38.1)	11,630 (39.7)	3.3
Race, n (%)			
White	15,218 (52.0)	14,812 (50.6)	2.8
Black	9,135 (31.2)	10,348 (35.4)	8.8
Hispanic	318 (1.1)	241 (0.8)	2.7
Other	1,981 (6.8)	1,073 (3.7)	14.0
Unknown	2,619 (8.9)	2,797 (9.6)	2.1
State, n (%)			
Missouri	8,238 (28.1)	8,203 (28.0)	0.3
New Jersey	6,553 (22.4)	6,788 (23.2)	1.9
Wisconsin	4,358 (14.9)	5,159 (17.6)	7.4
Iowa	3,859 (13.2)	3,908 (13.4)	0.5
Mississippi	3,642 (12.4)	2,575 (8.8)	11.9
Kansas	2,621 (9.0)	2,638 (9.0)	0.2
Insurance eligibility, n (%)			
Capitated	7,333 (25.1)	6,790 (23.2)	4.3
Dual Medicaid/Medicare	11,204 (38.3)	8,409 (28.7)	20.3
Year of index date, n (%)			
1998-2002	1,622 (5.5)	1,259 (4.3)	5.7
2003-2007	3,227 (11.0)	3,101 (10.6)	1.4
2008-2012	9,321 (31.8)	10,373 (35.4)	7.6
2013-2017	15,101 (51.6)	14,538 (49.7)	3.8
Quan-CCI, mean ± SD (median)	0.6 ± 1.3 (0.0)	0.5 ± 1.2 (0.0)	9.5
Unique mental health diagnoses, mean ± SD (median)	5.5 ± 4.8 (4.0)	1.0 ± 1.9 (0.0)	122.5
Pharmacy costs PPPY, mean ± SD (median), $US 2018	4,001 ± 8,467 (536)	1,124 ± 11,740 (6)	28.1
Medical costs PPPY, mean ± SD (median), $US 2018	21,314 ± 42,688 (7,205)	10,976 ± 37,614 (815)	25.7

CCI, Charlson comorbidity index; PPPY, per-patient-per-year; SD, standard deviation; US, United States.
[a]Patients with schizophrenia were matched to patients without schizophrenia 1:1 using exact matching factors (age groups) and propensity scores generated based on sex, race, state, insurance type (ie, capitated versus dual Medicaid/Medicare coverage), and year of the index date. Afterwards, the analysis was restricted to patients with schizophrenia aged 18-35 years old and their matched controls without schizophrenia.

HRU and Costs

- Young adults with schizophrenia had higher HRU relative to their matched pairs without schizophrenia during the follow-up period, including
 - 3.3 times more inpatient stays, 2.7 times more long-term care (LTC) stays, 2.6 times more emergency department visits, 1.9 times more outpatient visits, 17.0 times more mental health institute (MHI) stays, and 7.3 times more 1-day MHI visits (all $P < 0.001$; **Figure 2**)

Figure 2. Healthcare resource utilization during the 12-month follow-up period in young adults with versus without schizophrenia (N = 29,271 per cohort)[a]

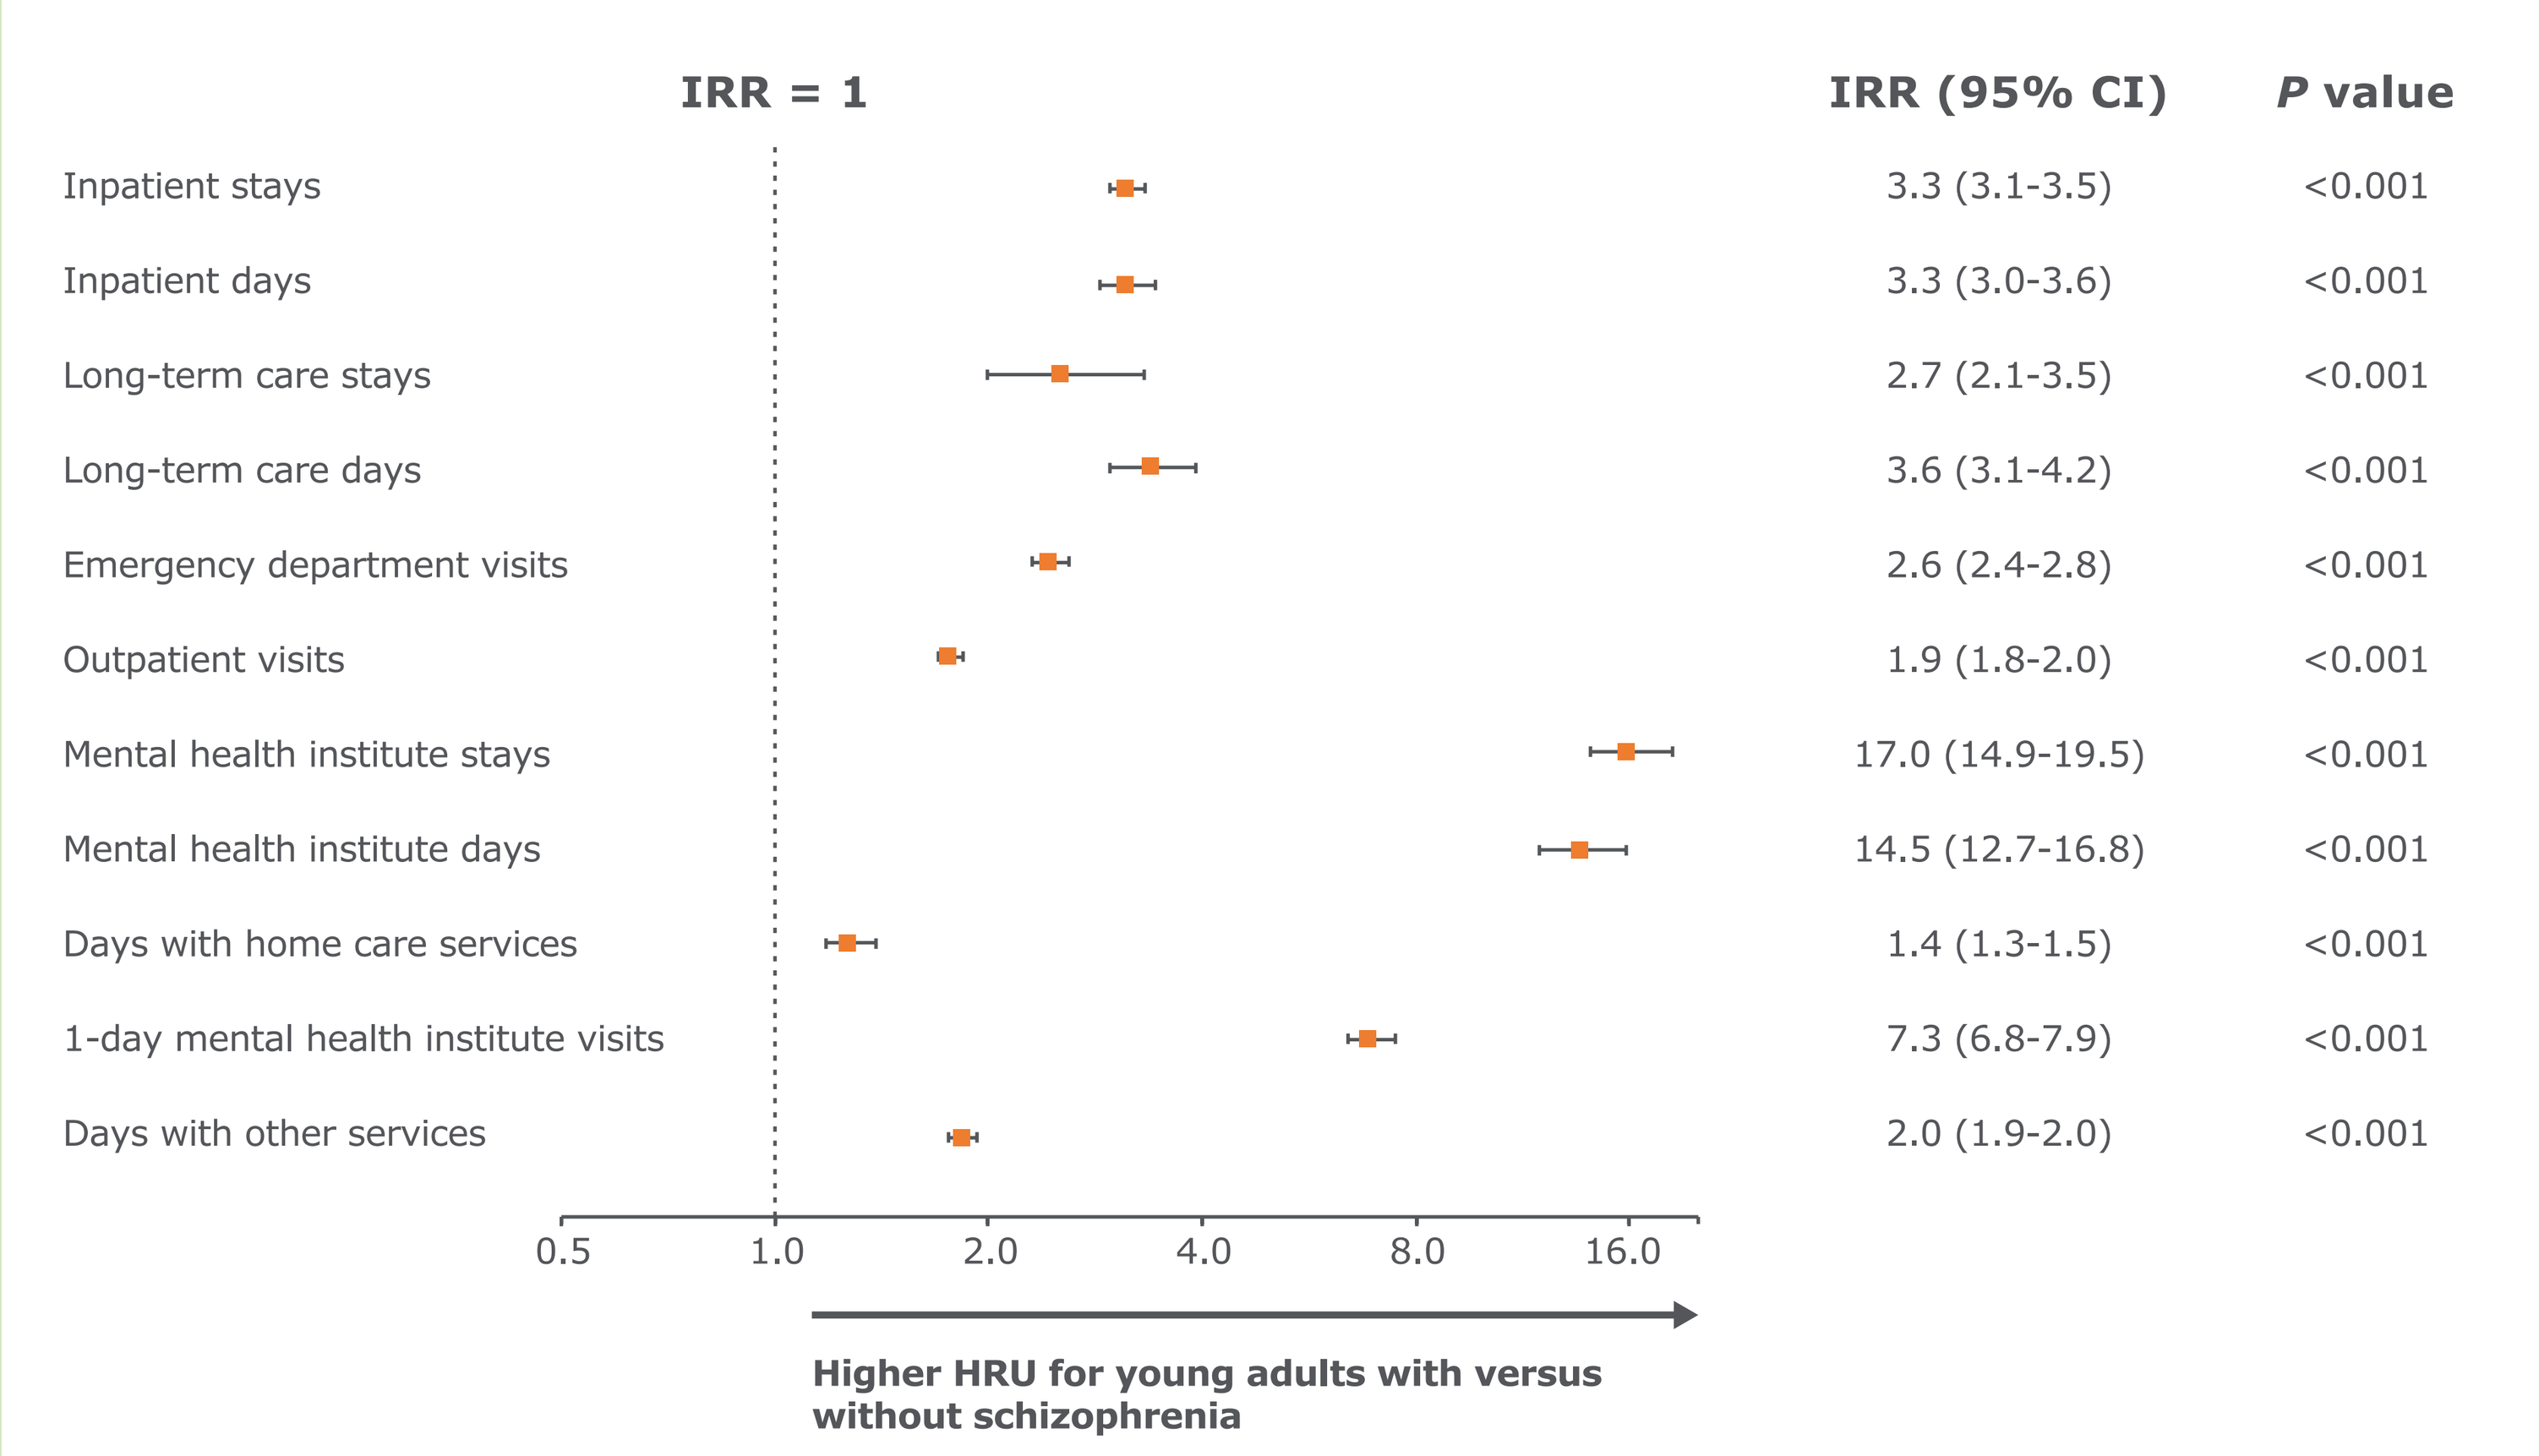
IRR = 1
IRR (95% CI)
P value
Inpatient stays
3.3 (3.1-3.5)
<0.001
Inpatient days
3.3 (3.0-3.6)
<0.001
Long-term care stays
2.7 (2.1-3.5)
<0.001
Long-term care days
3.6 (3.1-4.2)
<0.001
Emergency department visits
2.6 (2.4-2.8)
<0.001
Outpatient visits
1.9 (1.8-2.0)
<0.001
Mental health institute stays
17.0 (14.9-19.5)
<0.001
Mental health institute days
14.5 (12.7-16.8)
<0.001
Days with home care services
1.4 (1.3-1.5)
<0.001
1-day mental health institute visits
7.3 (6.8-7.9)
<0.001
Days with other services
2.0 (1.9-2.0)
<0.001
0.5
1.0
2.0
4.0
8.0
16.0
Higher HRU for young adults with versus without schizophrenia

CI, confidence interval; HRU, healthcare resource utilization; IRR, incidence rate ratio.
[a]HRU was compared between matched pairs using Poisson regression models adjusting for characteristics that remained unbalanced between matched pairs (ie, race, state, and insurance eligibility) to generate IRRs. Nonparametric bootstrap procedures were used to generate 95% CIs and *P* values.

- Young adults with schizophrenia incurred incremental total healthcare costs of $14,945 relative to their matched pairs without schizophrenia during the follow-up period ($P < 0.001$; **Figure 3**)
 - Incremental total costs were primarily driven by inpatient ($3,473), pharmacy ($2,609), and MHI stay costs ($2,216; all $P < 0.001$; **Figure 3**)
 - Incremental mental health–related costs ($13,276) accounted for 88.8% of the total incremental costs
- Young adults relative to all adult patients with schizophrenia had a comparable total incremental cost burden ($14,945 vs $14,087), but higher incremental inpatient costs ($3,743 vs $2,044) and lower incremental LTC costs ($1,108 vs $4,677)[6]

Figure 3. Healthcare costs during the 12-month follow-up period in young adults with versus without schizophrenia[a]

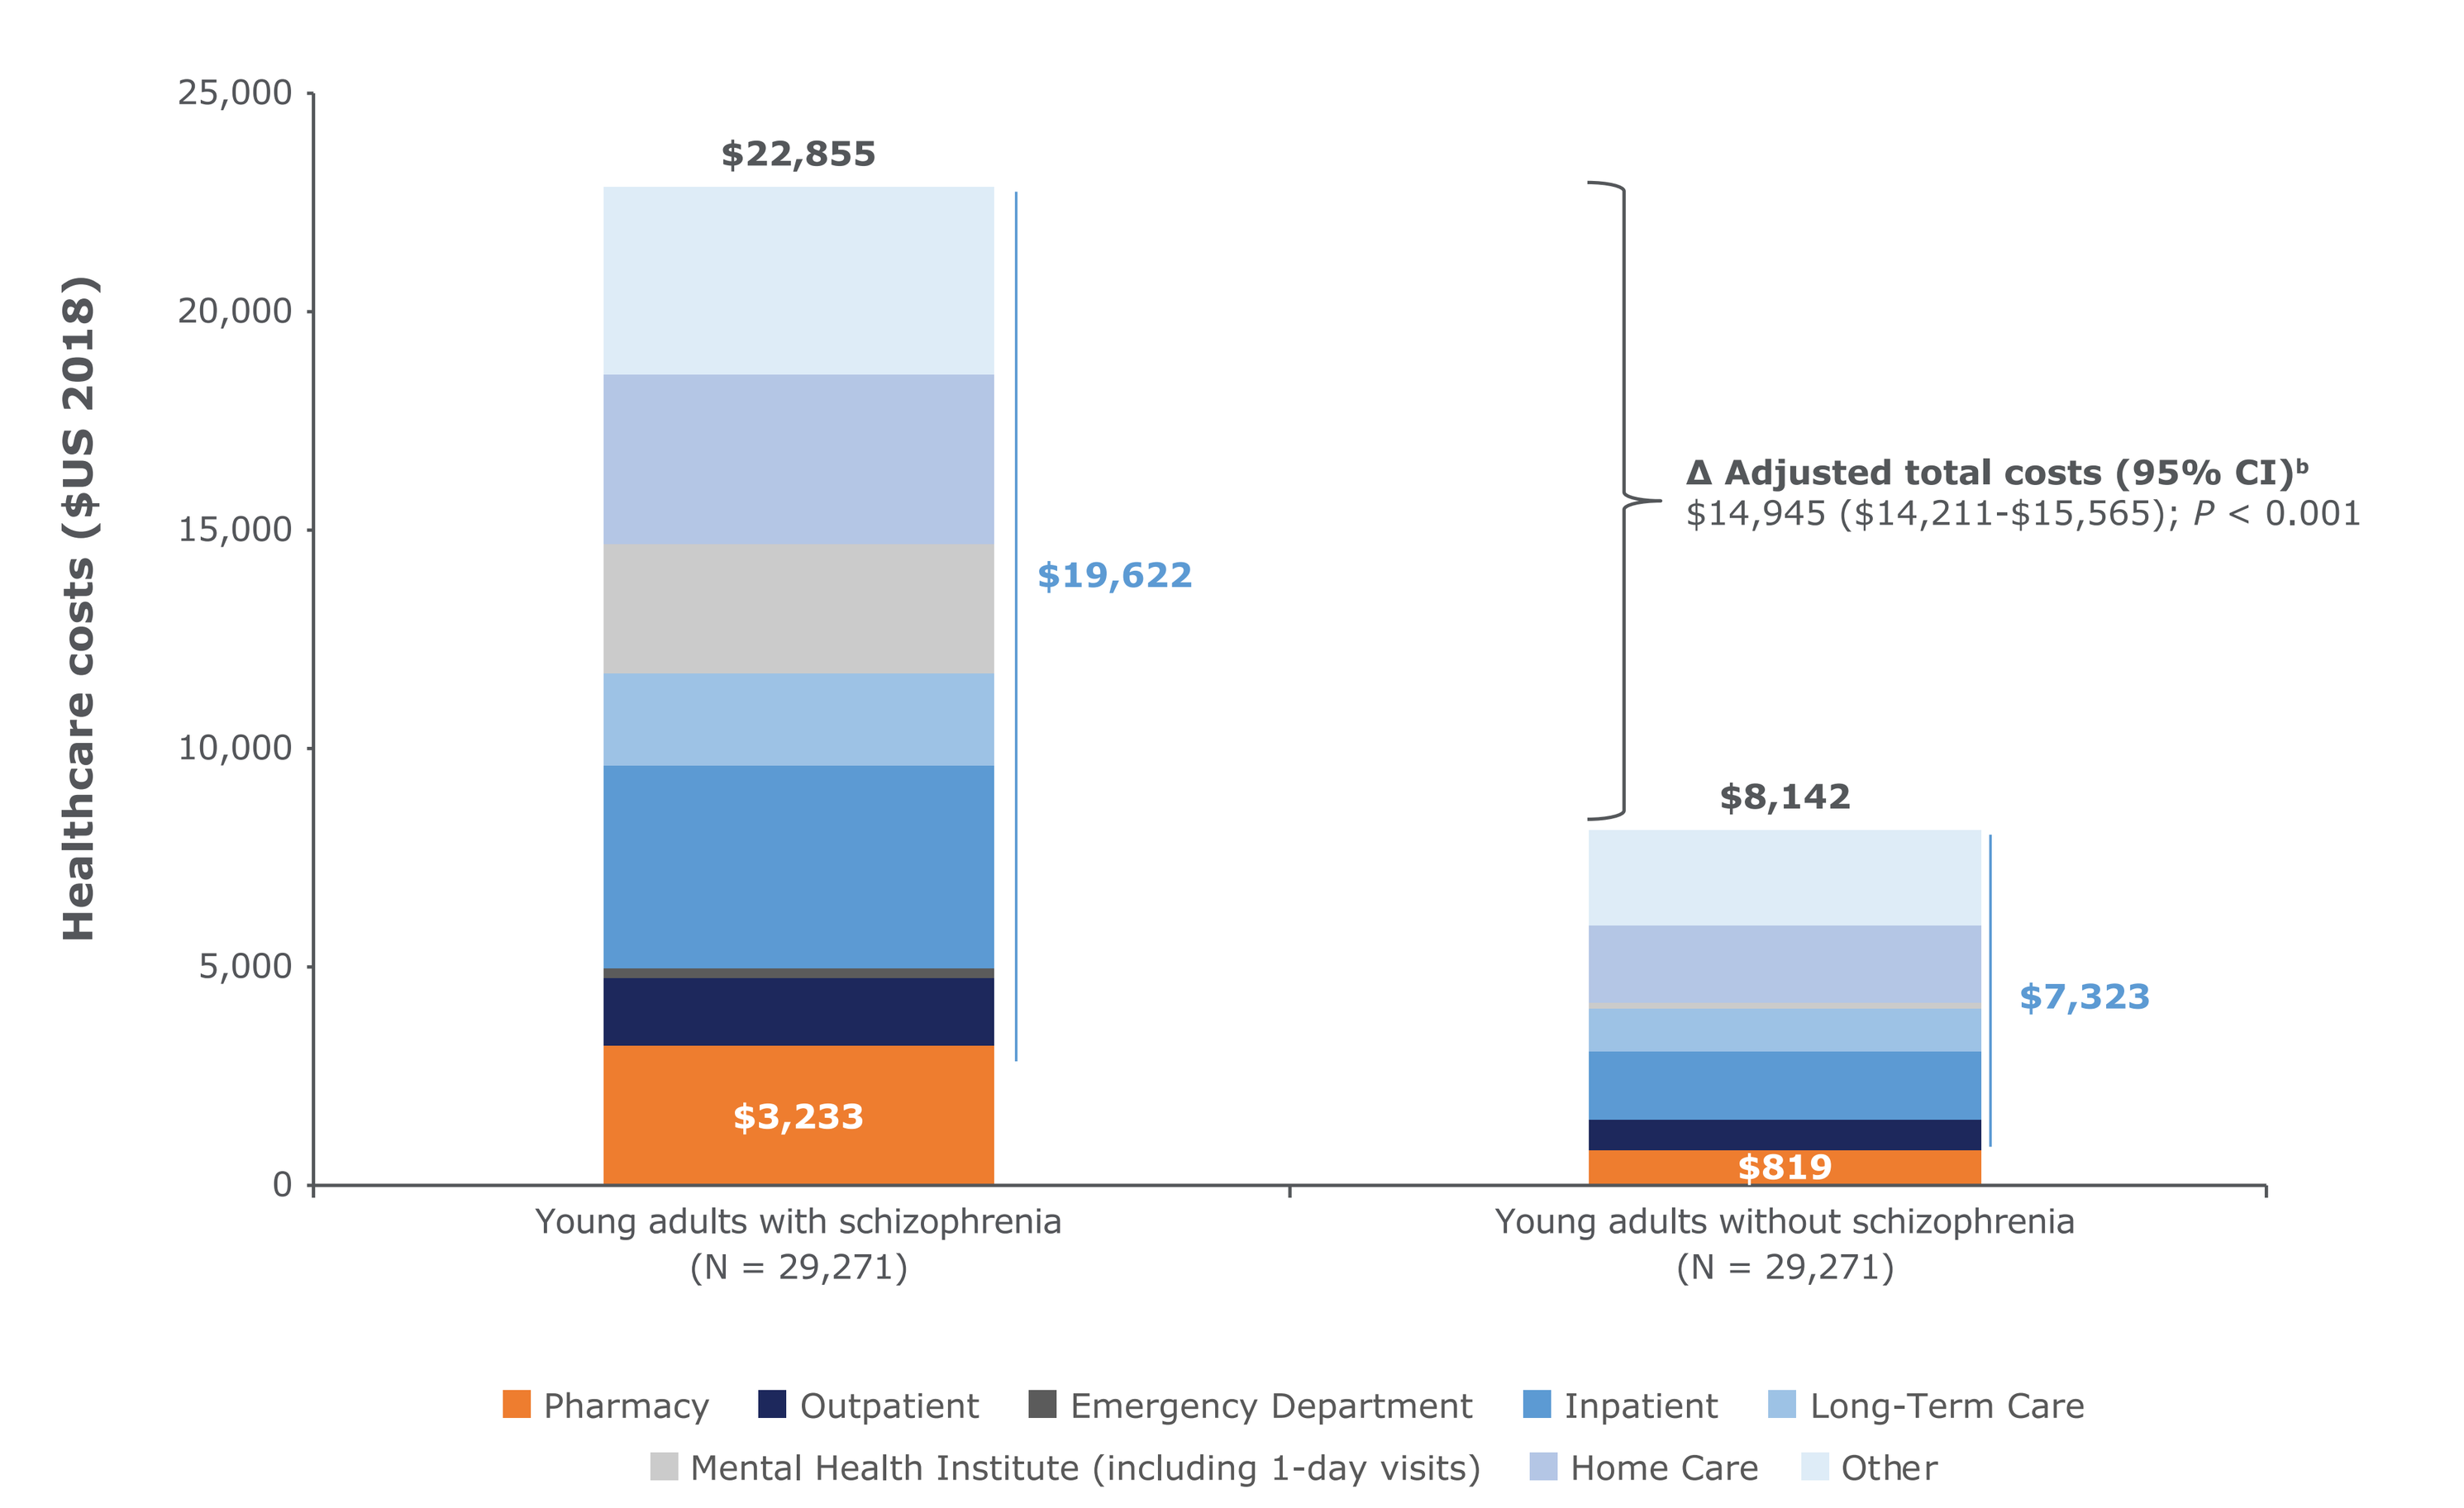
Healthcare costs ($US 2018)
25,000
20,000
15,000
10,000
5,000
0
$22,855
$19,622
$3,233
Δ Adjusted total costs (95% CI)[b]
$14,945 ($14,211-$15,565); P < 0.001
$8,142
$7,323
$819
Young adults with schizophrenia (N = 29,271)
Young adults without schizophrenia (N = 29,271)
Pharmacy
Outpatient
Emergency Department
Inpatient
Long-Term Care
Mental Health Institute (including 1-day visits)
Home Care
Other

CI, confidence interval; US, United States.
[a]Mean adjusted healthcare cost differences were estimated between matched pairs using ordinary least squares regression models adjusting for characteristics that remained unbalanced between matched pairs (ie, race, state, and insurance eligibility). Nonparametric bootstrap procedures were used to generate 95% CIs and *P* values.
[b]Mean adjusted cost differences between young adults with and without schizophrenia are not the same as raw mean cost differences due to adjustments made to account for race, state, and insurance eligibility.

LIMITATIONS

- Findings may not be representative of the Medicaid population with schizophrenia outside of the 6 states analyzed or generalizable to patients with schizophrenia without health insurance or with non-Medicaid insurance plans
- Analyses may be subject to residual confounding due to unmeasured confounders
- Analyses of administrative claims data depend on correct diagnosis, procedure, and drug codes, and coding inaccuracies may have led to case misidentification

CONCLUSIONS

- **In this 6-state study of Medicaid beneficiaries, young adults with schizophrenia incurred substantial incremental HRU and costs relative to young adults without schizophrenia**
- **The incremental cost burden of schizophrenia among young adults appeared comparable to that among the overall population of adults with schizophrenia aged 18 years and older**

References

1. National Alliance on Mental Illness. Schizophrenia. 2019. https://www.nami.org/learn-more/mental-health-conditions/schizophrenia. Accessed March 9, 2020.
2. National Institute of Mental Health. Schizophrenia Mental Health Information Statistics. 2019. https://www.nimh.nih.gov/health/statistics/schizophrenia.shtml. Accessed March 12, 2020.
3. Cloutier M et al. *J Clin Psychiatry*. 2016;77(6):764-771.
4. Huang A et al. *J Med Econ.* 2018;21(10):1026-1035.
5. Medicaid and CHIP Payment and Access Commission. Advising Congress on Medicaid and CHIP Policy. 2015. https://www.macpac.gov/wp-content/uploads/2015/06/June-2015-Report-to-Congress-on-Medicaid-and-CHIP.pdf. Accessed August 5, 2019.
6. Lafeuille MH et al. Poster presented at: Psych Congress; October 3-6, 2019; San Diego, CA.

Acknowledgments

This research was sponsored by Janssen Scientific Affairs, LLC.

Disclosures

M.-H. Lafeuille, D. Pilon, M. Zhdanava, A. Côté-Sergent, C. Rossi, and P. Lefebvre are employees of Analysis Group, Inc., a consulting company that has provided paid consulting services to Janssen Scientific Affairs, LLC, for the conduct of this study. C. Patel, D. Lin, and K. Joshi are employees of Janssen Scientific Affairs, LLC.

Supported by Janssen Scientific Affairs, LLC

Presented at the ISPOR Virtual 2020 Annual Meeting; May 18-20, 2020; Orlando, Florida