Economic Burden of Medicaid Beneficiaries with Schizophrenia not Adherent to Antipsychotics and with Schizophrenia Relapses

Dominic Pilon,[1] Charmi Patel,[2] Marie-Hélène Lafeuille,[1] Maryia Zhdanava,[1] Dee Lin,[2]
Aurélie Côté-Sergent,[1] Carmine Rossi,[1] Patrick Lefebvre,[1] Kruti Joshi[2]

[1]Analysis Group, Inc., Montréal, QC, Canada; [2]Janssen Scientific Affairs, LLC, Titusville, NJ, USA

BACKGROUND

- Schizophrenia is a chronic, debilitating mental illness estimated to affect about 1% of adults in the United States (US)[1] and approximately 2.9% of Medicaid beneficiaries[2]
- The economic burden associated with schizophrenia in the US was estimated in a 2013 analysis to exceed $155.7 billion annually[3]
- Antipsychotics (APs) are effective in reducing major symptoms of schizophrenia if patients remain adherent to their medications[4]
- However, many patients with schizophrenia have difficulty with long-term adherence to APs and experience detrimental health outcomes as a result, including relapses[4]
 - Use of second-generation long-acting injectables has been associated with improved adherence to and persistence on treatment compared with oral atypical APs[5]
- Among patients with schizophrenia, nonadherence to APs has been shown to be associated with incremental hospitalization costs[6]
- However, little is known about the economic burden of Medicaid beneficiaries with schizophrenia not adherent to APs and with prior relapses relative to general population

OBJECTIVES

- To evaluate incremental healthcare resource utilization (HRU) and costs of Medicaid patients with schizophrenia not adherent to APs and with prior schizophrenia relapses relative to those of patients without schizophrenia

METHODS

Data Source

- Medicaid data from 6 states were used: Iowa (Q1 1998 to Q3 2016), Kansas (Q1 2001 to Q1 2018), Mississippi (Q1 2006 to Q1 2018), Missouri (Q1 1997 to Q1 2018), New Jersey (Q1 1997 to Q1 2014), and Wisconsin (Q1 2004 to Q4 2013)
- Information on Medicaid enrollment, demographic variables, as well as claims for medical visits and prescription drugs, including corresponding costs, was available

Study Design

- A retrospective matched cohort design was used
 - Patients with schizophrenia were matched to patients without schizophrenia
 - Afterwards, the analysis was restricted to patients with schizophrenia who were not adherent to APs (proportion of days covered by APs ≤80%) and had ≥2 schizophrenia-related hospitalizations (proxies for relapses) in the 12-month pre-index period, as well as their matched controls without schizophrenia
- The index date was the last observed diagnosis of schizophrenia (in the cohort with schizophrenia) or the last service claim (in the control cohort) with ≥12 months of continuous Medicaid enrollment before and after it
- Baseline characteristics were described in the 12-month pre-index (ie, baseline) period; HRU and costs were described and compared during the 12-month post-index (ie, follow-up) period

Inclusion Criteria

- Cohort with schizophrenia
 - ≥2 claims on different days with a diagnosis of schizophrenia (International Classification of Diseases, 9th Revision, Clinical Modification [ICD-9-CM] code: 295.xx; International Classification of Diseases, 10th Revision, Clinical Modification [ICD-10-CM]: F20.xx, F21, F25.xx)
 - ≥1 claim with a diagnosis of schizophrenia with ≥12 months of continuous Medicaid enrollment before and after the claim, with the latest claim being the index date
 - Proportion of days covered by APs ≤80% during the baseline period
 - ≥2 schizophrenia-related hospitalizations during the baseline period
- Control cohort
 - No claims with a diagnosis of schizophrenia during the continuous Medicaid enrollment period
 - ≥1 service claim with ≥12 months of continuous Medicaid enrollment before and after the claim, with the latest claim being the index date
- Both cohorts
 - Aged ≥18 years as of the index date

Figure 1. Identification of the study population

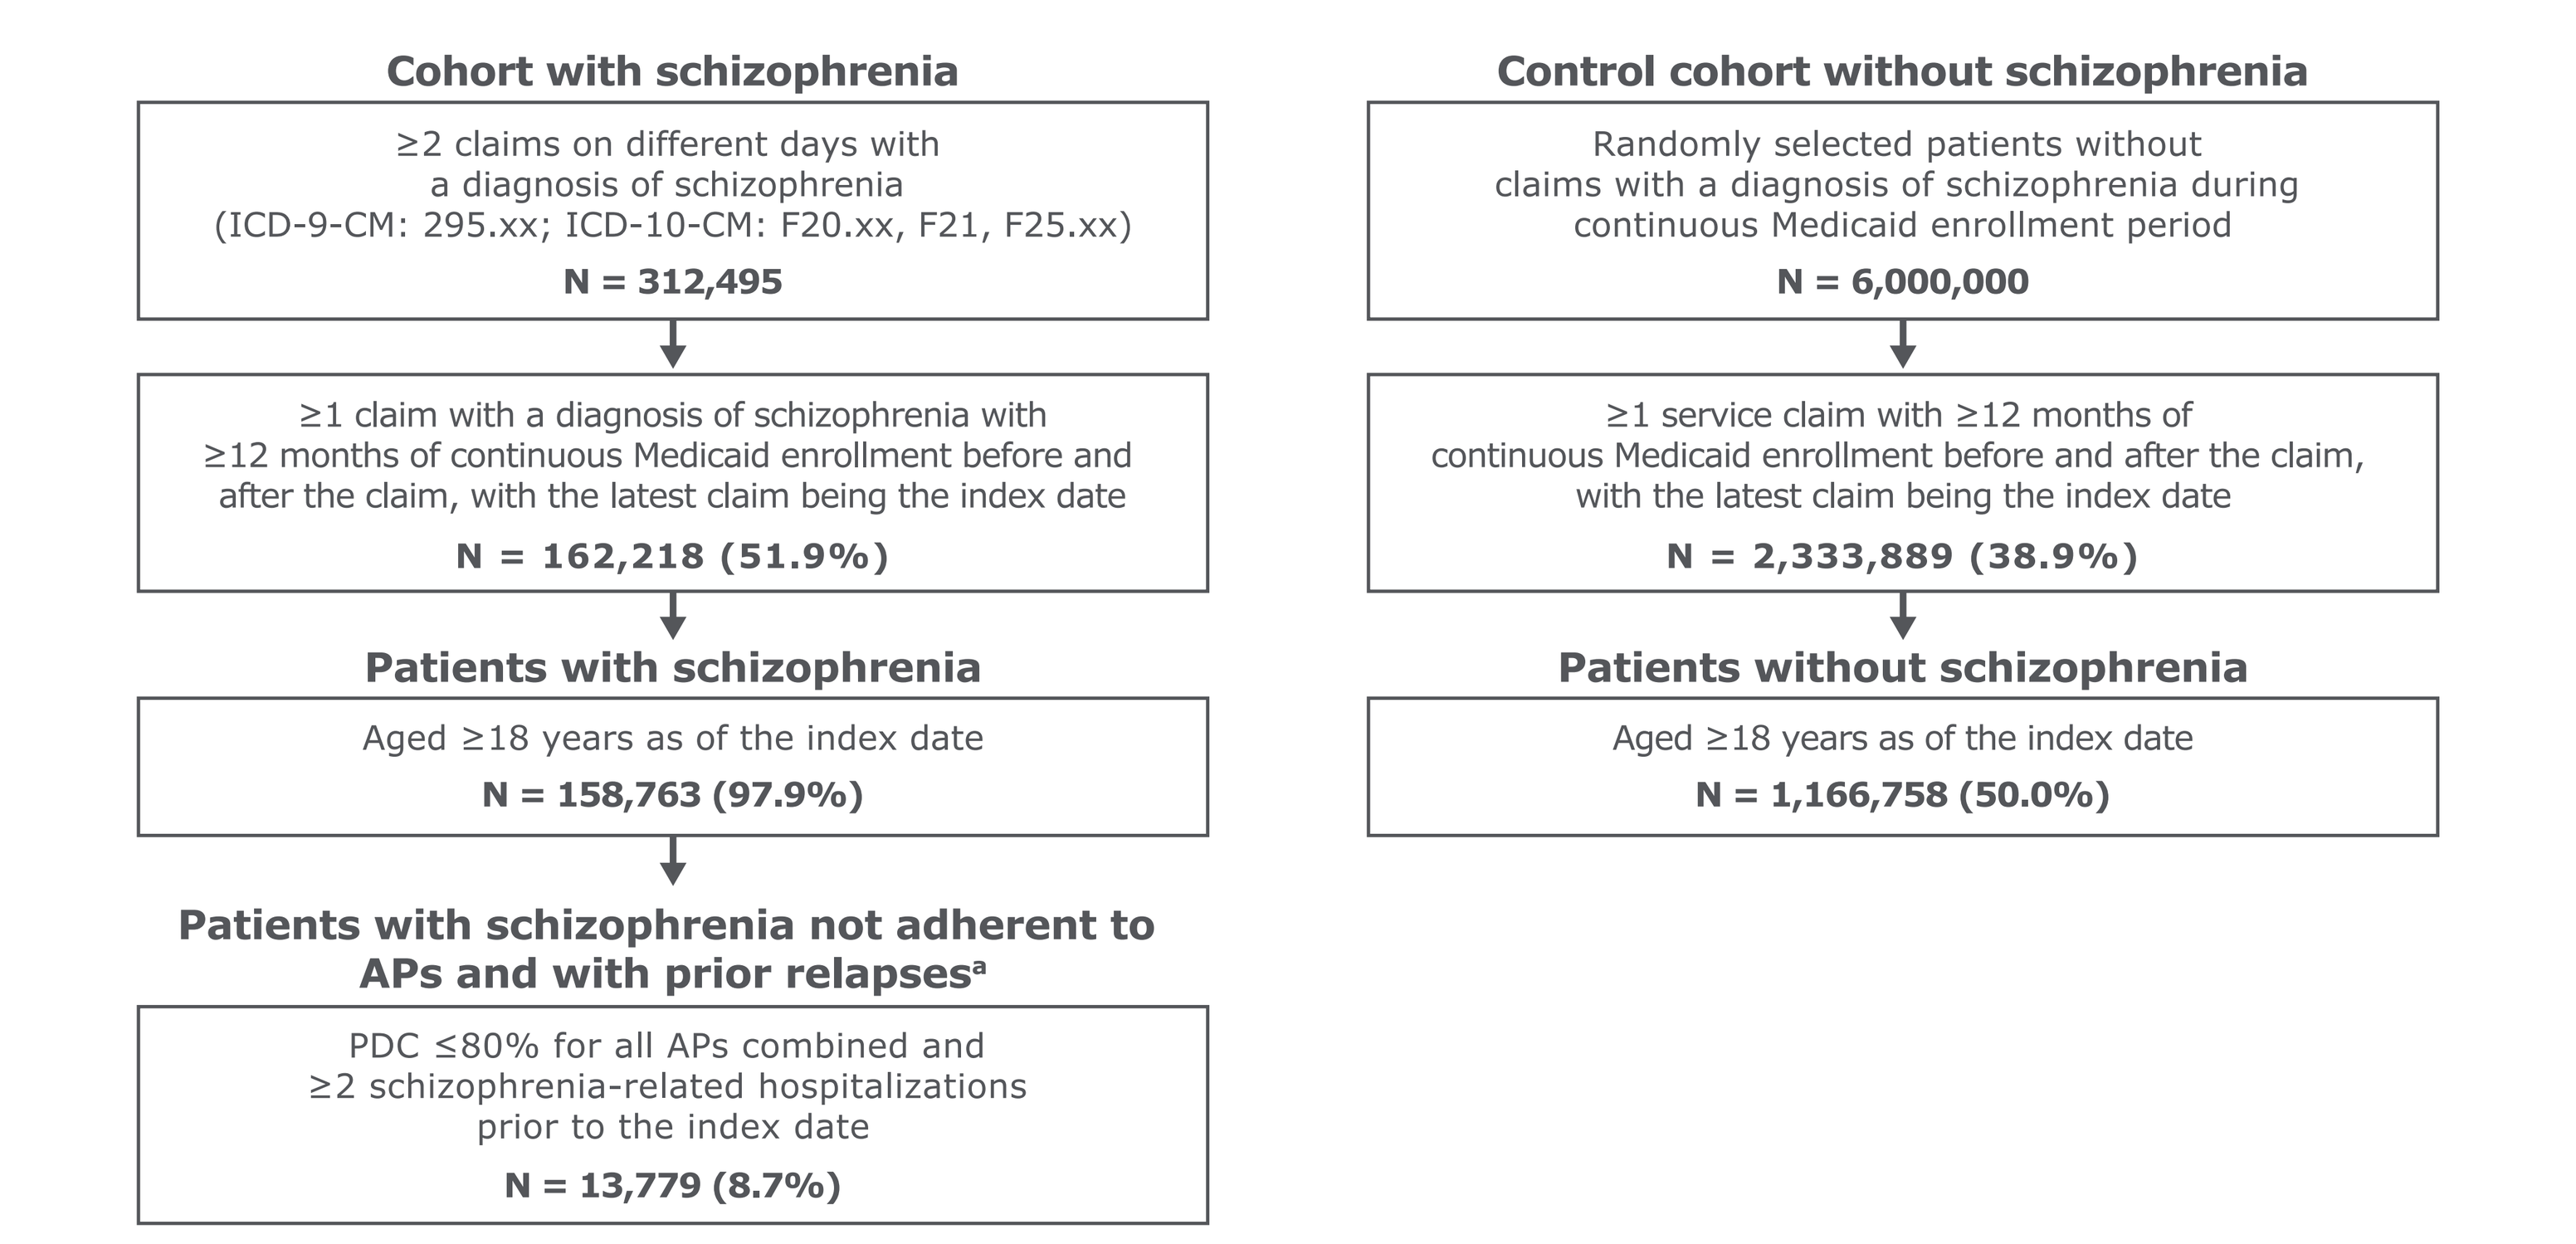
Cohort with schizophrenia
≥2 claims on different days with a diagnosis of schizophrenia (ICD-9-CM: 295.xx; ICD-10-CM: F20.xx, F21, F25.xx)
N = 312,495
≥1 claim with a diagnosis of schizophrenia with ≥12 months of continuous Medicaid enrollment before and after the claim, with the latest claim being the index date
N = 162,218 (51.9%)
Patients with schizophrenia
Aged ≥18 years as of the index date
N = 158,763 (97.9%)
Patients with schizophrenia not adherent to APs and with prior relapses[a]
PDC ≤80% for all APs combined and ≥2 schizophrenia-related hospitalizations prior to the index date
N = 13,779 (8.7%)
Control cohort without schizophrenia
Randomly selected patients without claims with a diagnosis of schizophrenia during continuous Medicaid enrollment period
N = 6,000,000
≥1 service claim with ≥12 months of continuous Medicaid enrollment before and after the claim, with the latest claim being the index date
N = 2,333,889 (38.9%)
Patients without schizophrenia
Aged ≥18 years as of the index date
N = 1,166,758 (50.0%)

AP, antipsychotic; ICD-CM-9, International Classification of Diseases, 9th Revision, Clinical Modification; ICD-10-CM, International Classification of Diseases, 10th Revision, Clinical Modification; PDC, proportion of days covered.
[a]Patients with schizophrenia were matched to patients without schizophrenia. The analysis was then restricted to patients with schizophrenia not adherent to APs and with relapses during the baseline period and their matched schizophrenia-free controls.

Outcome Measures

- HRU and healthcare costs were reported per-patient-per year over the 12-month follow-up period based on medical and prescription drug claims
- Costs were adjusted for inflation using the medical care component of the US Consumer Price Index and were reported in 2018 US dollars

Statistical Analysis

- Patients with and without schizophrenia were matched 1:1 using exact matching factors (ie, age group) and propensity scores generated based on sex, race, state, insurance eligibility (ie, capitated versus dual Medicaid/Medicare coverage), and year of the index date
- The analysis was then restricted to patients with schizophrenia not adherent to APs and with relapses during the baseline period and to their matched schizophrenia-free controls
- HRU was compared between matched pairs using Poisson regression models, and nonparametric bootstrap procedures were used to generate 95% confidence intervals (CIs) and *P* values
- Mean cost differences between matched pairs were estimated using ordinary least squares regression models, and nonparametric bootstrap procedures were used to generate 95% CIs and *P* values
- The comparisons of HRU and costs were adjusted for characteristics that remained unbalanced between the matched pairs (ie, race, state, and insurance eligibility)

RESULTS

Study Sample

- Among 158,763 patients with schizophrenia, 13,779 (8.7%) were not adherent to APs and had experienced prior relapses (**Figure 1**); their baseline characteristics are presented in **Table 1**

Table 1. Baseline characteristics in patients with schizophrenia not adherent to APs and with prior relapses and their matched pairs without schizophrenia[a]

	Patients With Schizophrenia Not Adherent to APs and With Prior Relapses N = 13,779	Patients Without Schizophrenia N = 13,779	Standardized Difference (%)
Age at index date, mean ± SD (median), years	47.1 ± 15.4 (47.2)	48.0 ± 17.4 (47.4)	5.3
Female, n (%)	6,619 (48.0)	6,300 (45.7)	4.6
Race, n (%)			
White	7,435 (54.0)	8,120 (58.9)	10.0
Black	4,747 (34.5)	4,332 (31.4)	6.4
Hispanic	62 (0.4)	65 (0.5)	0.3
Other	981 (7.1)	743 (5.4)	7.1
Unknown	554 (4.0)	519 (3.8)	1.3
State, n (%)			
Missouri	5,590 (40.6)	5,187 (37.6)	6.0
New Jersey	4,912 (35.6)	4,146 (30.1)	11.9
Mississippi	1,222 (8.9)	1,644 (11.9)	10.0
Iowa	772 (5.6)	885 (6.4)	3.5
Wisconsin	758 (5.5)	1,297 (9.4)	14.9
Kansas	525 (3.8)	620 (4.5)	3.5
Insurance eligibility, n (%)			
Capitated	1,228 (8.9)	2,488 (18.1)	27.0
Dual Medicaid/Medicare	6,683 (48.5)	6,931 (50.3)	3.6
Year of index date, n (%)			
1998-2002	1,538 (11.2)	957 (6.9)	14.7
2003-2007	2,803 (20.3)	1,961 (14.2)	16.2
2008-2012	4,053 (29.4)	3,991 (29.0)	1.0
2013-2017	5,385 (39.1)	6,870 (49.9)	21.8
Quan-CCI, mean ± SD (median)	2.2 ± 2.6 (1.0)	1.5 ± 2.3 (1.0)	27.2
Unique mental health diagnoses, mean ± SD (median)	8.5 ± 5.4 (7.0)	1.2 ± 1.9 (0.0)	179.5
Pharmacy costs PPPY, mean ± SD (median), US $2018	5,478 ± 9,713 (2,429)	2,516 ± 10,259 (62)	29.6
Medical costs PPPY, mean ± SD (median), US $2018	39,958 ± 50,467 (24,155)	16,739 ± 42,039 (1,879)	50.0

CCI, Charlson Comorbidity Index; PPPY, per-patient-per-year; SD, standard deviation; US, United States.
[a]Patients with schizophrenia were matched to patients without schizophrenia 1:1 using exact matching factors (age groups) and propensity scores generated based on sex, race, state, insurance eligibility (ie, capitated vs dual Medicaid/Medicare coverage), and year of the index date. Afterwards, the analysis was restricted to patients with schizophrenia not adherent to APs and with prior relapses and their matched controls without schizophrenia.

HRU and Costs

- Patients with schizophrenia not adherent to APs and with prior relapses had higher HRU relative to their matched pairs without schizophrenia during the follow-up period, including
 - 3.1 times as many inpatient stays, 2.1 times as many long-term care (LTC) stays, 2.6 times as many emergency department visits, 1.5 times as many outpatient visits, 14.6 times as many mental health institute (MHI) stays, and 4.9 times as many 1-day MHI visits (all *P* < 0.001; **Figure 2**)

Figure 2. Healthcare resource utilization during the 12-month follow-up period in patients with schizophrenia not adherent to APs and with prior relapses versus patients without schizophrenia (N = 13,779 per cohort)[a]

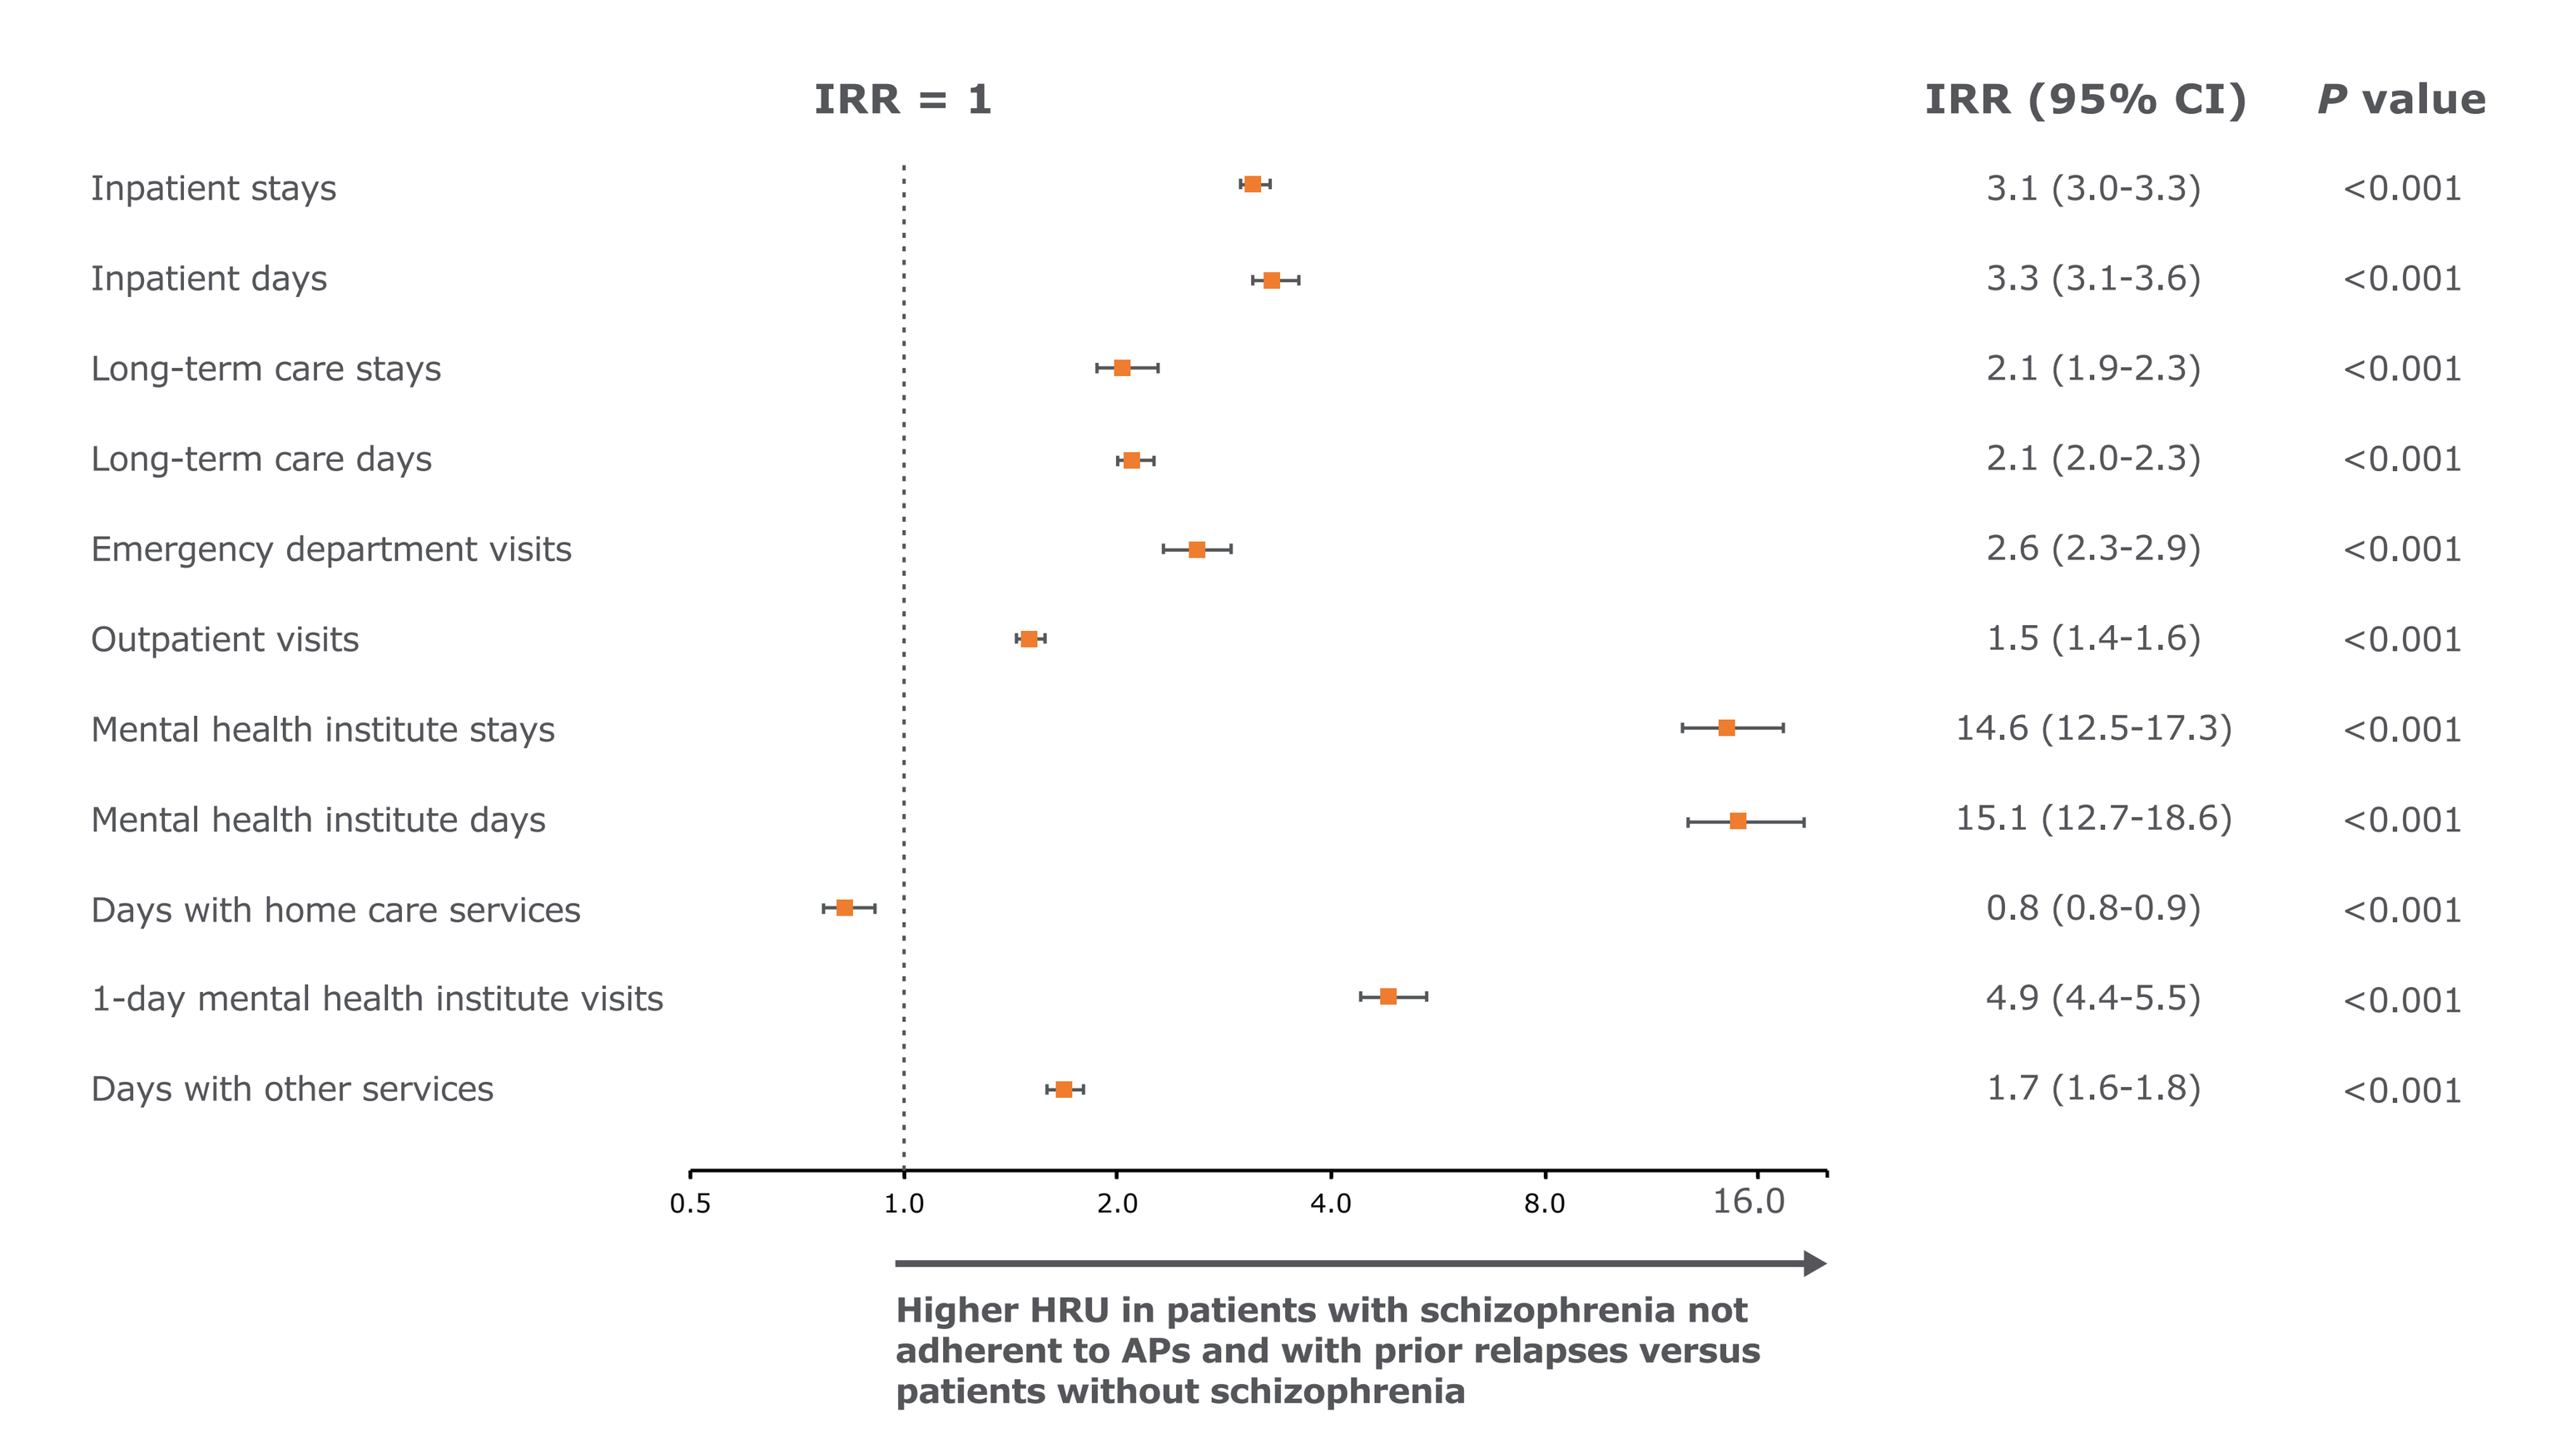
IRR = 1
IRR (95% CI)
P value
Inpatient stays
3.1 (3.0-3.3)
<0.001
Inpatient days
3.3 (3.1-3.6)
<0.001
Long-term care stays
2.1 (1.9-2.3)
<0.001
Long-term care days
2.1 (2.0-2.3)
<0.001
Emergency department visits
2.6 (2.3-2.9)
<0.001
Outpatient visits
1.5 (1.4-1.6)
<0.001
Mental health institute stays
14.6 (12.5-17.3)
<0.001
Mental health institute days
15.1 (12.7-18.6)
<0.001
Days with home care services
0.8 (0.8-0.9)
<0.001
1-day mental health institute visits
4.9 (4.4-5.5)
<0.001
Days with other services
1.7 (1.6-1.8)
<0.001
0.5
1.0
2.0
4.0
8.0
16.0
Higher HRU in patients with schizophrenia not adherent to APs and with prior relapses versus patients without schizophrenia

CI, confidence interval; HRU, healthcare resource utilization; IRR, incidence rate ratio.
[a]HRU was compared between matched pairs using Poisson regression models adjusting for characteristics that remained unbalanced between matched pairs (ie, race, state, and insurance eligibility) to generate IRRs. Nonparametric bootstrap procedures were used to generate 95% CIs and *P* values.

- Patients with schizophrenia not adherent to APs and with prior relapses incurred incremental total healthcare costs of $21,688 relative to their matched pairs without schizophrenia during the follow-up period (*P* < 0.001; **Figure 3**)
- Among patients with schizophrenia not adherent to APs and with prior relapses:
 - Incremental total costs were primarily driven by inpatient ($8,417), LTC ($5,427), pharmacy ($2,547), and MHI stay ($2,503) costs (all *P* < 0.001; **Figure 3**)
 - Incremental mental health-related costs ($16,331) accounted for 75.3% of the total incremental costs
- Patients not adherent to APs and with prior relapses, relative to the overall population of patients with schizophrenia, had a greater incremental cost burden ($21,688 vs $14,087)[7]

Figure 3. Healthcare costs during the 12-month follow-up period in patients with schizophrenia not adherent to APs and with prior relapses versus patients without schizophrenia[a]

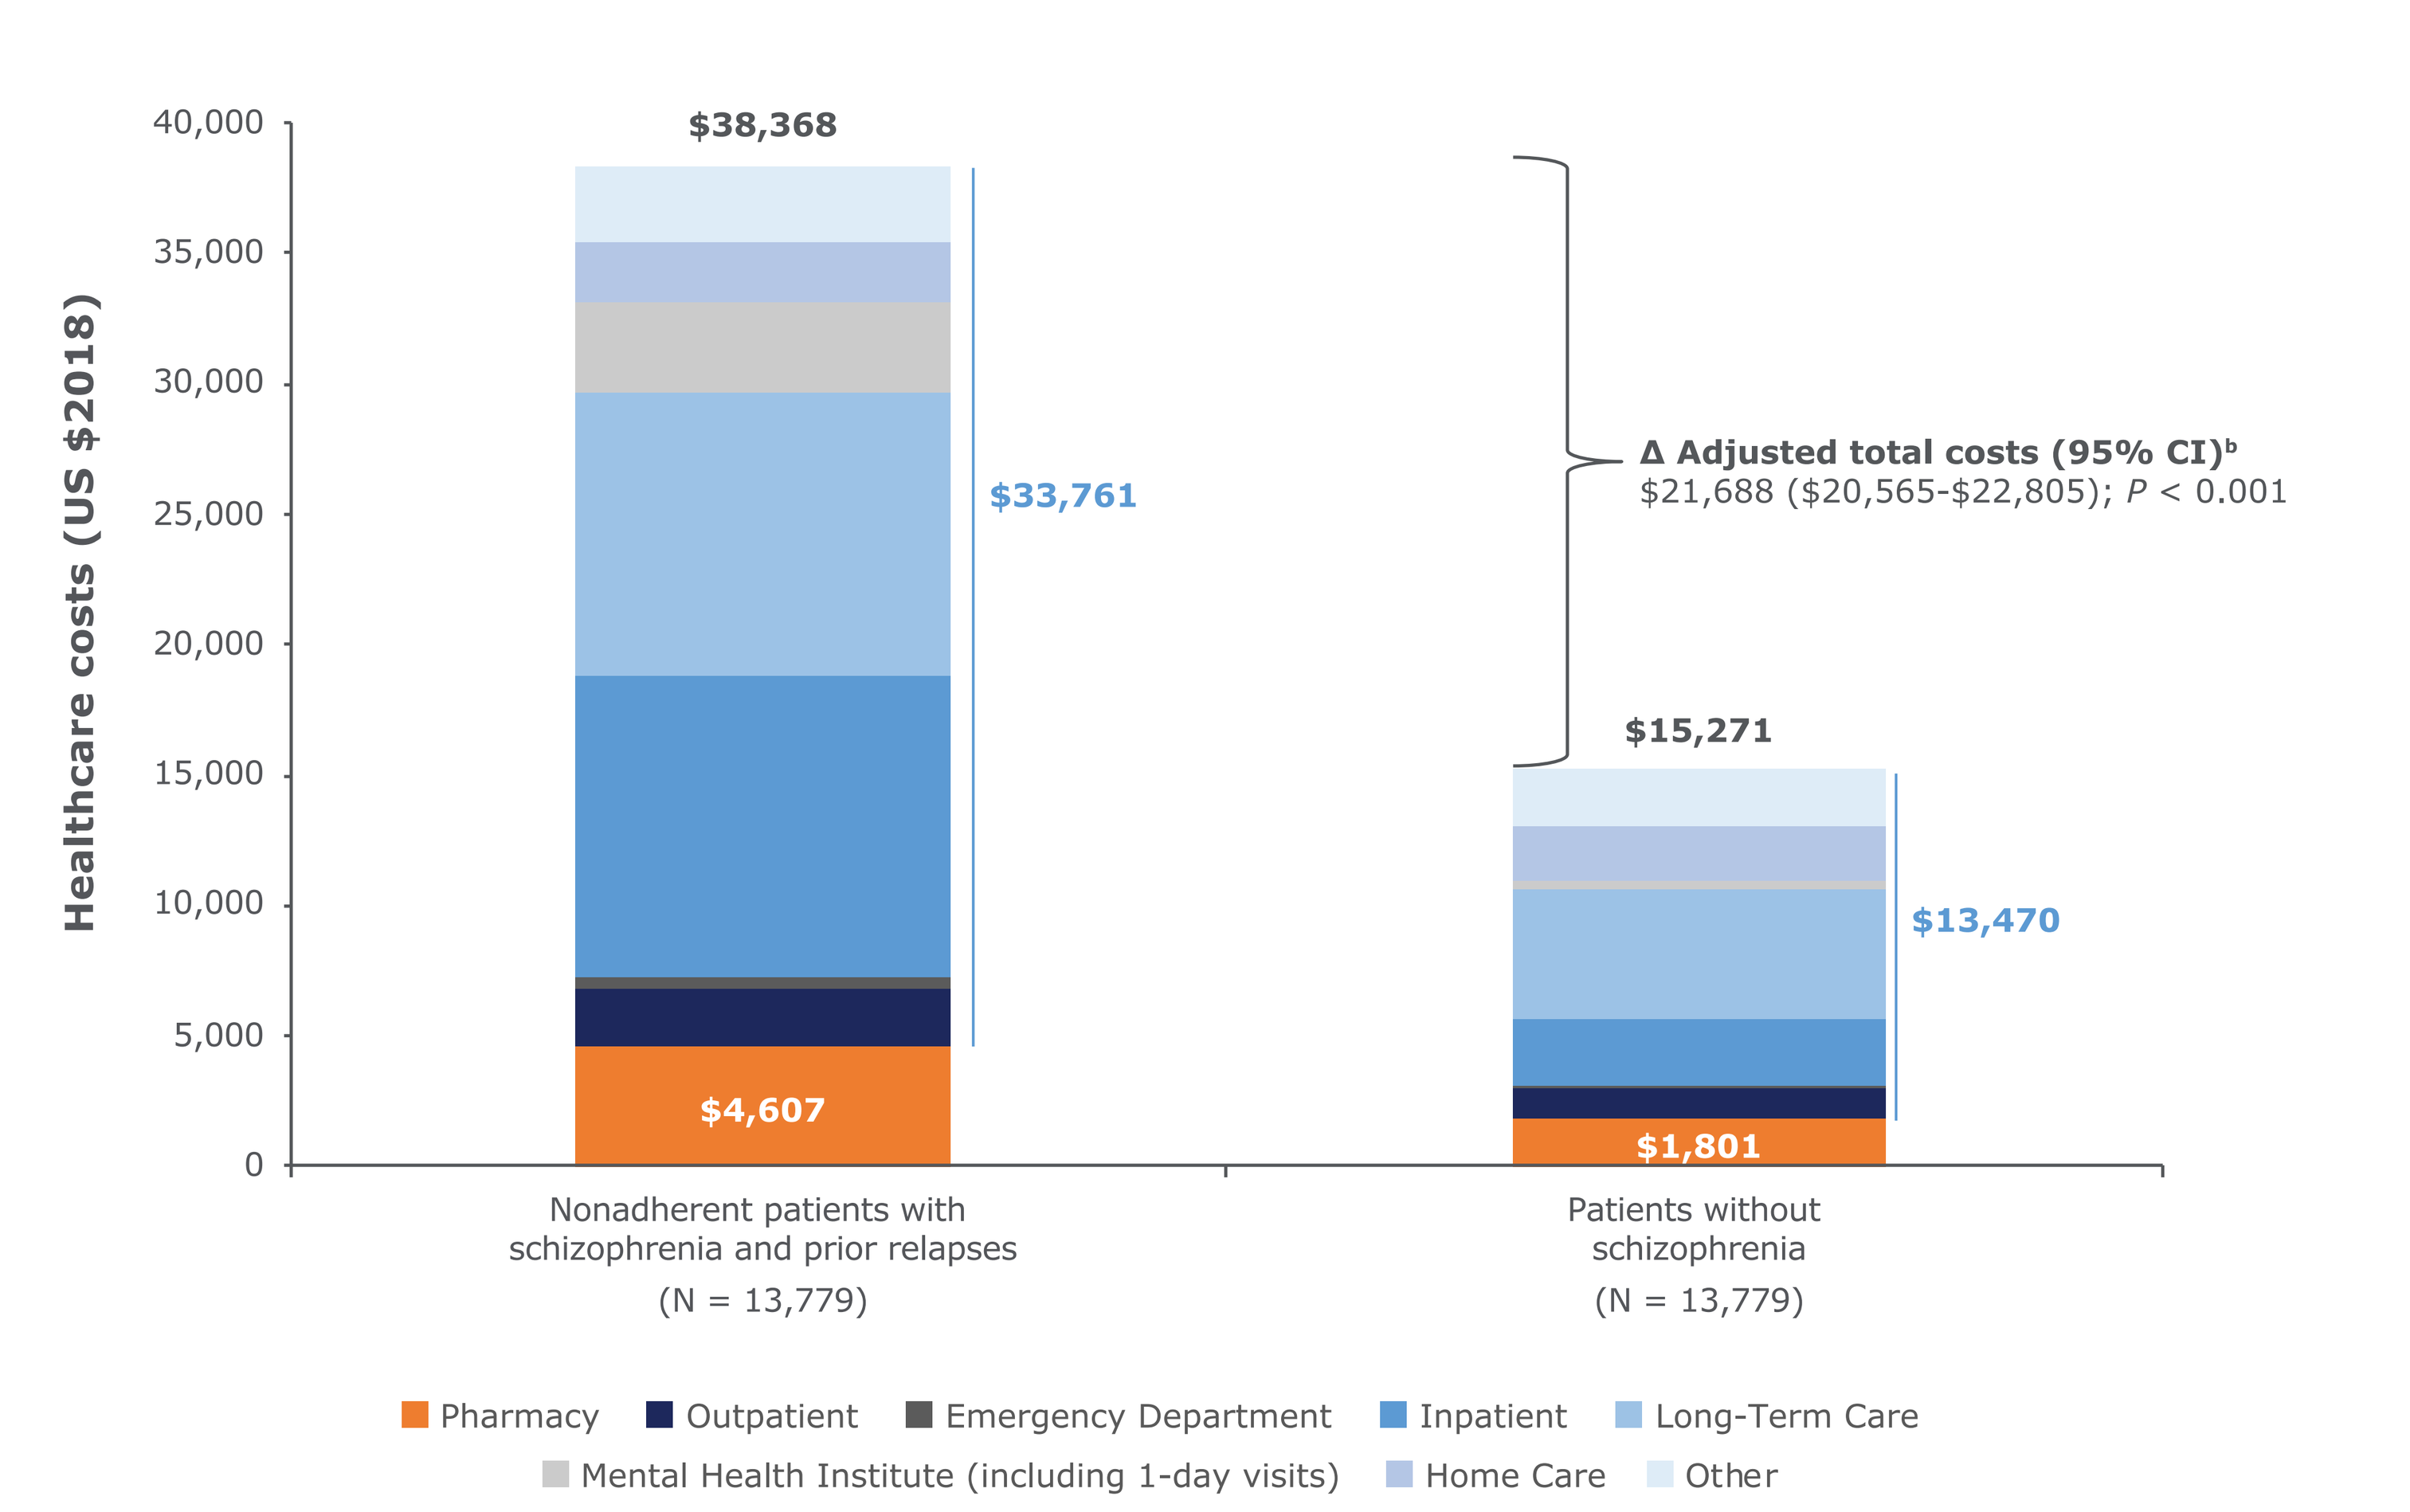
Healthcare costs (US $2018)
40,000
35,000
30,000
25,000
20,000
15,000
10,000
5,000
0
$38,368
$33,761
$4,607
$15,271
$13,470
$1,801
Δ Adjusted total costs (95% CI)[b]
$21,688 ($20,565-$22,805); P < 0.001
Nonadherent patients with schizophrenia and prior relapses
(N = 13,779)
Patients without schizophrenia
(N = 13,779)
Pharmacy
Outpatient
Emergency Department
Inpatient
Long-Term Care
Mental Health Institute (including 1-day visits)
Home Care
Other

CI, confidence interval; US, United States.
[a]Mean adjusted healthcare cost differences were estimated between matched pairs using ordinary least squares regression models adjusting for characteristics that remained unbalanced between matched pairs (ie, race, state, and insurance eligibility). Nonparametric bootstrap procedures were used to generate 95% CIs and *P* values.
[b]Mean adjusted cost differences between nonadherent patients with schizophrenia and prior relapses and patients without schizophrenia are not the same as raw mean cost differences due to adjustments made to account for race, state, and insurance eligibility.

LIMITATIONS

- Findings may not be representative of the Medicaid population with schizophrenia outside of the 6 states analyzed or generalizable to patients with schizophrenia without health insurance or with non-Medicaid insurance plans
- Analyses may be subject to residual confounding due to unmeasured confounders
- Analyses of administrative claims data depended on correct diagnosis, procedure, and drug codes, and coding inaccuracies may have led to case misidentification
- Patients with 2 or more schizophrenia-related hospitalizations may have experienced multiple distinct relapses or a single relapse episode encompassing several hospitalizations and spanning several weeks

CONCLUSIONS

- **Medicaid beneficiaries with schizophrenia not adherent to APs and with prior relapses incurred substantial incremental HRU and costs relative to those without schizophrenia**
- **The incremental cost burden among patients with schizophrenia not adherent to APs and with prior relapses appeared higher than that of the overall population of patients with schizophrenia**

References

1. National Institute of Mental Health (NIMH). Schizophrenia Mental Health Information Statistics. 2019. Available at https://www.nimh.nih.gov/health/statistics/schizophrenia.shtml. Accessed March 12, 2020.
2. Medicaid and CHIP Payment and Access Commission. 2015. https://www.macpac.gov/wp-content/uploads/2015/06/June-2015-Report-to-Congress-on-Medicaid-and-CHIP.pdf. Accessed August 5, 2019.
3. Cloutier M et al. *J Clin Psychiatry*. 2016;77(6):764-771.
4. Haddad PM et al. *Patient Relat Outcome Meas*. 2014;5:43-62.
5. Pilon D et al. *Clin Ther*. 2017;39(10):1972-1985.
6. Pennington M et al. *Pharmacoeconomics*. 2018;36:941-955.
7. Lafeuille MH et al. Presented at Psych Congress; October 3-6, 2019; San Diego, CA.

Acknowledgments

This research was sponsored by Janssen Scientific Affairs, LLC.

Disclosures

D. Pilon, M-H. Lafeuille, M. Zhdanava, A. Côté-Sergent, C. Rossi, and P. Lefebvre are employees of Analysis Group, Inc., a consulting company that has provided paid consulting services to Janssen Scientific Affairs, LLC, for the conduct of this study. C. Patel, D. Lin, and K. Joshi are employees of Janssen Scientific Affairs, LLC.

Supported by Janssen Scientific Affairs, LLC

Presented at the Virtual ISPOR 2020 Conference; May 18-20, 2020; Orlando, Florida